CG-806, a First-in-Class FLT3/BTK Inhibitor, and Venetoclax Synergize to Inhibit Cell Proliferation and to Induce Apoptosis in Aggressive B-cell Lymphomas

Nasrin Rastgoo[1], Matthew Thayer[1], Khalid Benbatoul[1], Stephen Howell[2], William Rice[1], Hongying Zhang[1]
[1]Aptose Biosciences Inc, San Diego, CA; [2]UC San Diego Moores Cancer Center, San Diego, CA

APTOSE BIOSCIENCES

AACR 2020, abstract # 4417

Abstract

Double/triple-hit lymphomas (DHL/THL, harboring concurrent MYC, BCL2 and/or BCL6 rearrangements) and double-expressor lymphomas (DEL, with MYC/BCL2 co-overexpression without underlying rearrangements) account for 7-12% and 20-30% of diffuse large B-cell lymphomas (DLBCLs), respectively, and have poor outcome after standard/intensive immunochemotherapy (Petrich, 2014; Smith, 2018). More effective and less toxic treatments for these aggressive B-cell lymphomas represent a significant unmet medical need. Venetoclax, a BCL2 inhibitor, significantly inhibits the proliferation of DLBCL cells in culture but has less promising clinical efficacy as a single agent. CG-806, a FLT3/BTK inhibitor, exhibits potent antitumor activity against cell lines and primary cells over a range of hematologic malignancies by suppressing the driver and compensatory pathways (inhibits phosphorylation of BTK, FLT3, PDGFRα, SYK, SRC, ERK, MAPK, STAT5 and AKT, and decreases MYC level). CG-806 is currently in a Phase I a/b trial in patients with CLL/SLL or non-Hodgkin's lymphomas (NCT03893682). We sought to investigate the antitumor effect of CG-806 alone or in combination with venetoclax on DHL/THL/DEL cells using MTS based proliferation assay, flow cytometry assays of apoptosis and cell cycle, and immunoblotting.

In VAL, SU-DHL6 and DOHH2 DHL/THL cells that overexpress MYC and BCL2, CG-806 inhibited their proliferation with IC_{50}s of 0.07, 0.13 and 0.005 µM, respectively. CG-806 was 5-20X more potent than venetoclax and exposure to CG-806 significantly enhanced sensitivity to venetoclax. SU-DHL2, a DHL cell line that does not overexpress MYC or BCL2, was inhibited more effectively by CG-806 than venetoclax (IC_{50} 0.9 vs 6.3 µM, respectively). In the DEL cell line U2932, CG-806 and venetoclax had similar potency as single agents (IC_{50} 0.7 µM); when combined, the IC_{50} was markedly improved to 0.05 µM. Meanwhile, combination of CG-806 with venetoclax enhanced induction of apoptosis in DHL/THL/DEL cells in a concentration and time-dependent manner as detected by annexin V staining and cleavage of PARP and caspase 3. Recent studies ascribed the resistance of DLBCLs to venetoclax to upregulation of MCL1 and MAPK signaling. Our study showed that CG-806 inhibited expression of the anti-apoptotic protein Mcl-1, overcame the effect of venetoclax-induced Mcl-1 and increased the level of proapoptotic BIM. While venetoclax did not affect MAPK signaling, CG-806 alone and in combination with venetoclax reduced p-MEK1/2, p-JNK and MYC in a cell line dependent manner.

In conclusion, CG-806 inhibits driver and rescue pathways to directly and potently kill the aggressive B-cell lymphomas including DHL/THL/DEL and enhances the proapoptotic effect of venetoclax, thereby highlighting CG-806 as a promising candidate for the treatment of patients harboring unfavorable BCL2/MYC/BCL6 translocations and/or overexpression and supporting clinical development of CG-806 in patients with B-cell malignancies intolerant, resistant, or refractory to venetoclax.

Materials and Methods

- Cytotoxicity assay: Cell viability of cultured cell lines was measured by MTS assay and expressed as percent vehicle control.
- Apoptosis: assayed via flow cytometry with annexin V / PI staining
- Cell proliferation: CellTrace™ CFSE Cell Proliferation Kit was used for in vivo labeling of cells and to trace multiple generations using dye dilution by flow cytometry.
- Caspase 3/7 activity: CellEvent™ Caspase-3/7 Green Detection Reagent and SYTOX™ AADvanced™ dead cell stain was used for detection of activated caspases 3 and 7 in apoptotic cells.
- Immunoblotting – Whole cell lysates were collected from cells after treatment with either Vehicle (DMSO) or CG-806 alone or in combination with venetoclax at the concentrations and for the times listed in each figure. The total and phosphorylated levels of the indicated proteins were measured by immunoblotting.

Disclosures: N. Rastgoo: Aptose BioSciences Inc. Employment; M. Thayer: Aptose Biosciences Inc. Employment; K. Benbatoul: Aptose Biosciences Inc. Employment; S. Howell: Membership on an entity's Board of Directors or advisory committees, Aptose Bioscience Inc.; W. Rice: Equity Ownership and Patents & Royalties, Aptose Biosciences Inc.; H. Zhang: Aptose Biosciences Inc. Employment.

CG-806 Potently Kills Double-Hit and Double-Expressor Lymphoma Cells and Synergizes with Venetoclax

		DOHH2	VAL	SUDHL6	U2932	SUDHL2
CG-806	IC_{50} (µM)	0.005	0.07	0.13	0.70	0.9
Venetoclax	IC_{50} (µM)	0.012	0.03	2.30	0.70	6.3
CG-806 + Venetoclax	IC_{50} (µM)	0.0008	0.01	0.02	0.06	**
	CI*	< 0.5	< 0.5	< 0.5	< 0.5	**

*Combination Index (CI) was calculated by CompuSyn software; CI< 0.5 is considered synergistic effect.
**Study is ongoing and data are pending.

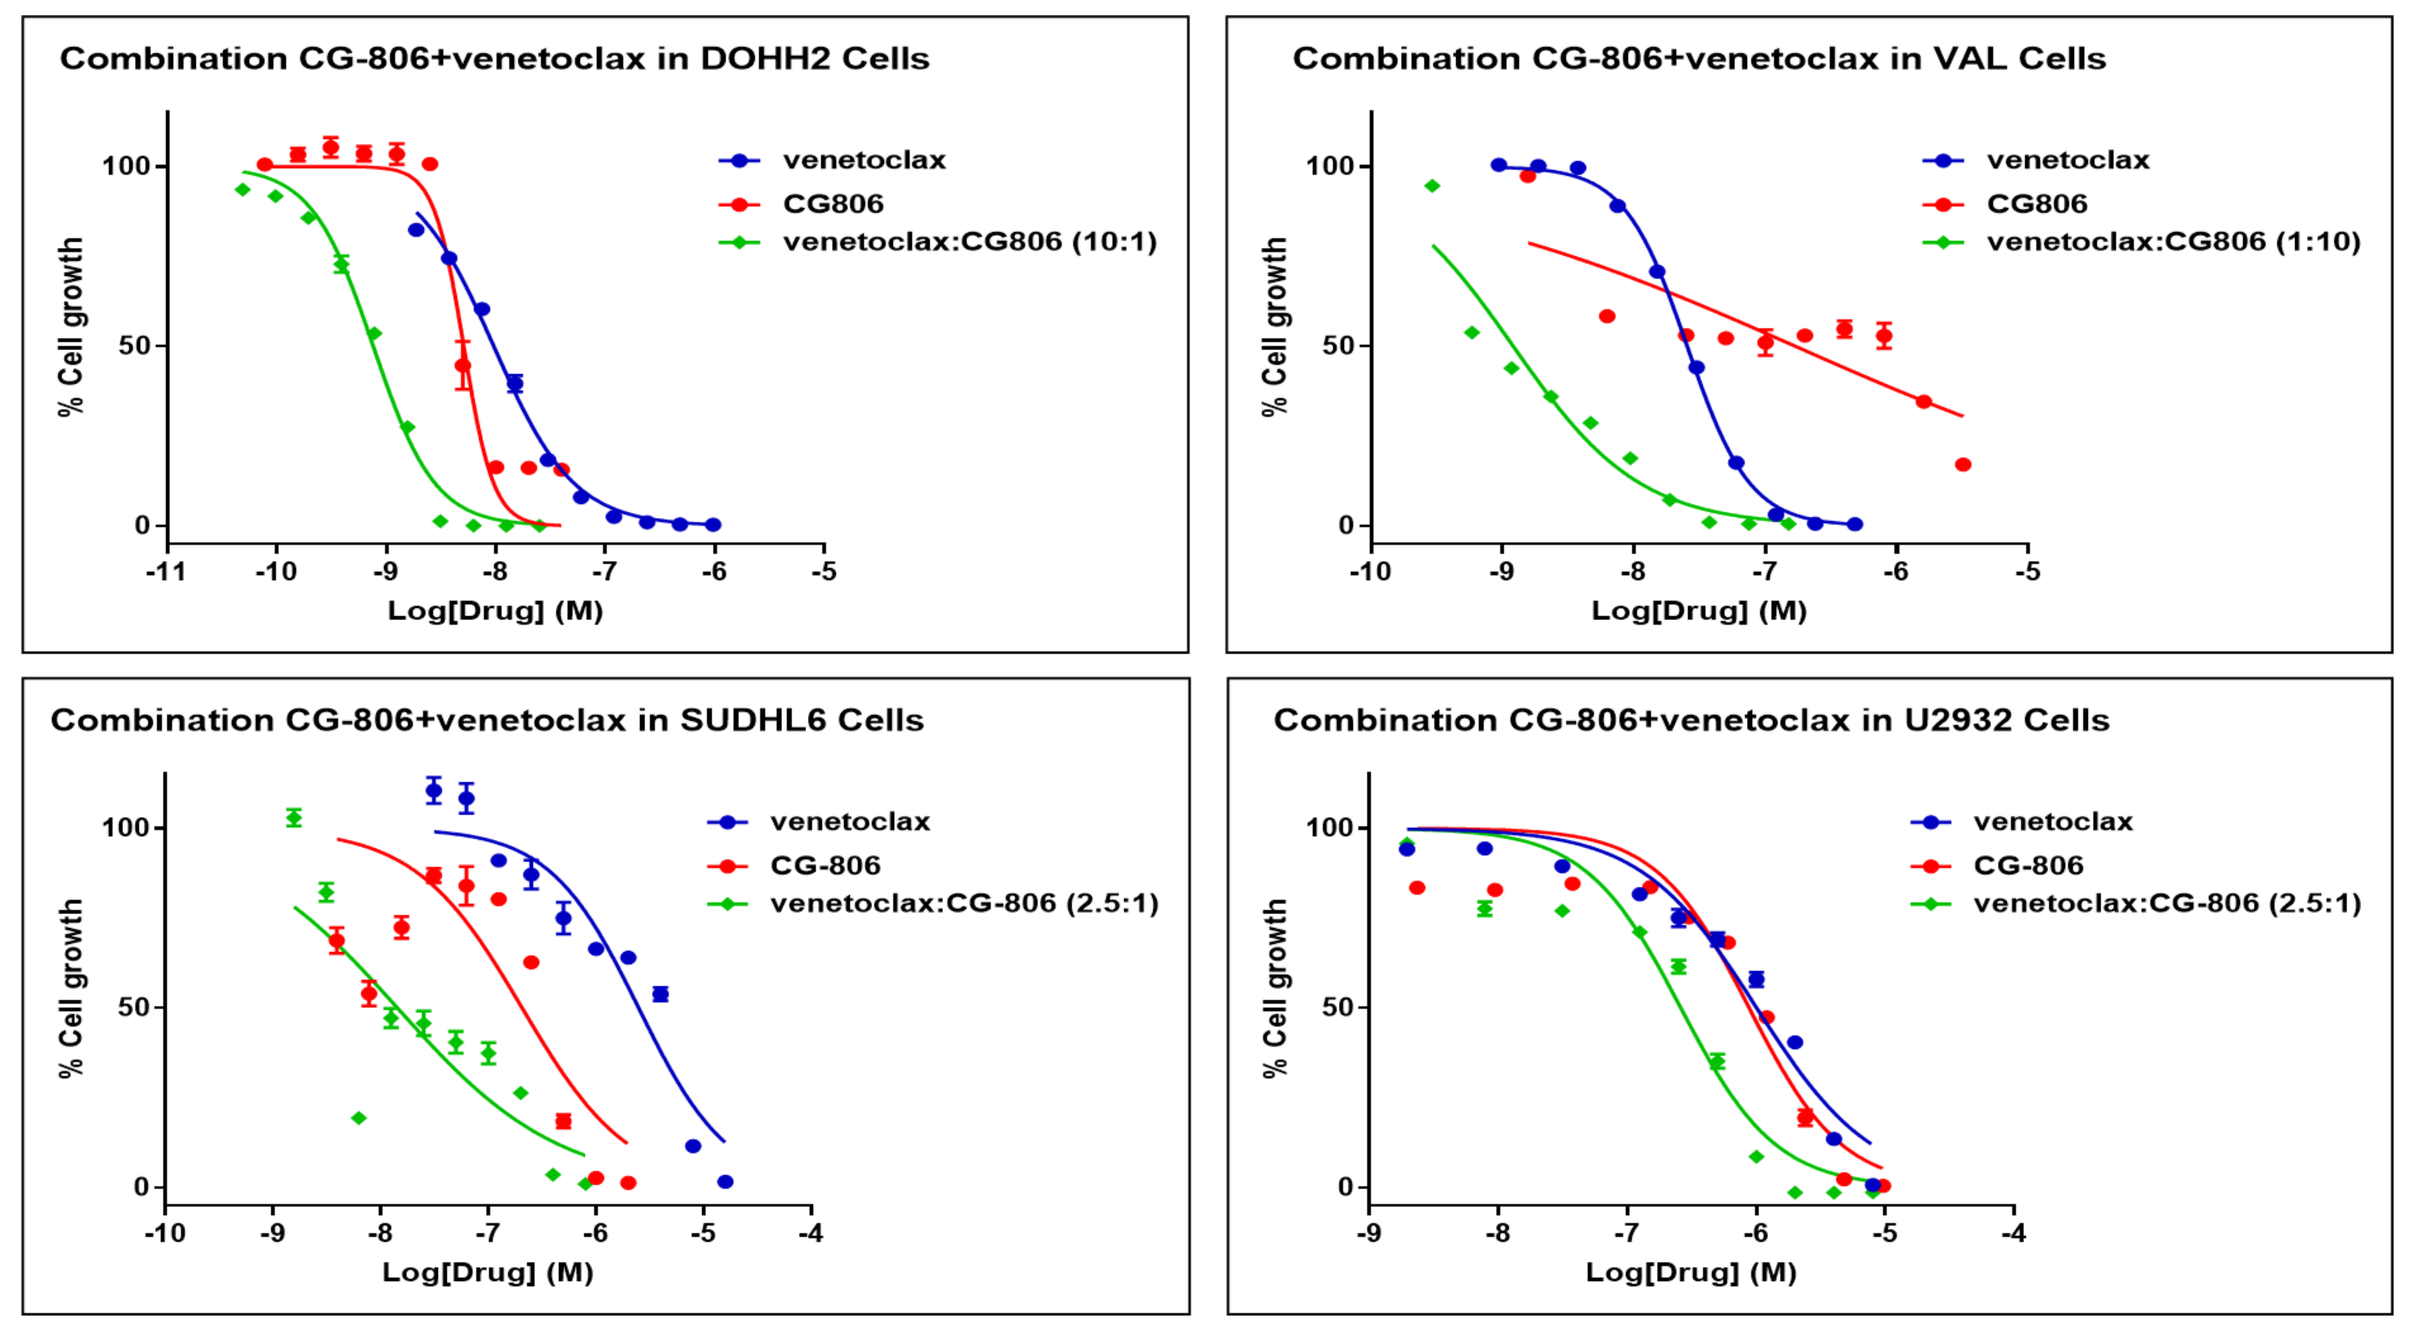
Combination CG-806+venetoclax in DOHH2 Cells
Combination CG-806+venetoclax in VAL Cells
Combination CG-806+venetoclax in SUDHL6 Cells
Combination CG-806+venetoclax in U2932 Cells
venetoclax
CG806
venetoclax:CG806 (10:1)
venetoclax:CG806 (1:10)
CG-806
venetoclax:CG-806 (2.5:1)
% Cell growth
Log[Drug] (M)

Survival of DHL and DEL cells as a function of concentration for CG-806 and Venetoclax individually and in combination. Cells were exposed to drugs for 72 h and then subjected to MTS assay. IC_{50}s were calculated by GraphPad Prism software.

DHL/DEL	Cell line	Original Diagnosis	Characterization
DHL	DOHH2	Folicular centroblastic lymphoma	MYC and BCL2 rearrangements
	SUDHL2	ABC-like DLBCL	MYC and BCL2 rearrangements
	SUDHL6	GCB-like DLBCL	MYC and BCL2 rearrangements
	VAL	B-acute lymphoblastic Leukemia	MYC and BCL2 and BCL6 rearrangements
DEL	U2932	ABC-like DLBCL	MYC and BCL2 overexpression, BCL6 expression

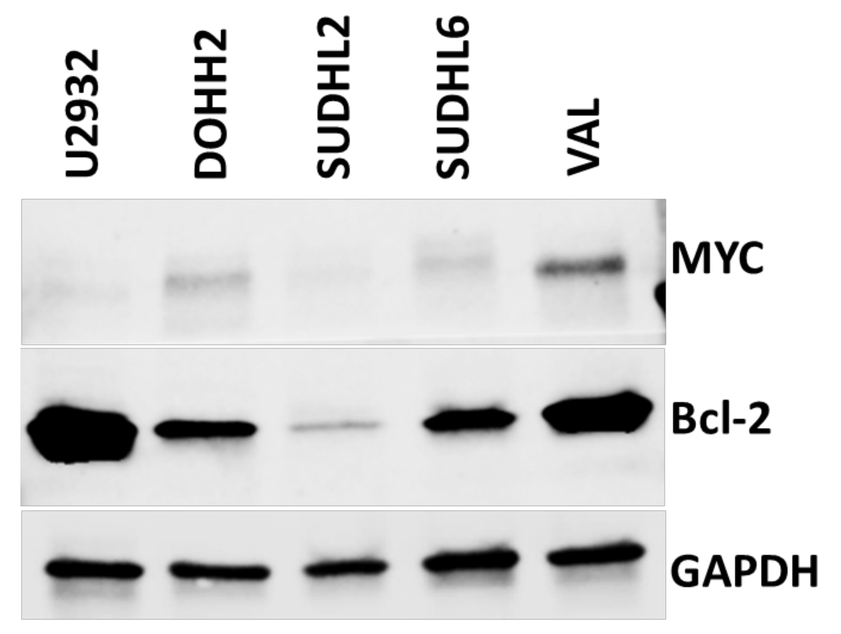
U2932
DOHH2
SUDHL2
SUDHL6
VAL
MYC
Bcl-2
GAPDH

Characteristics of the double-hit lymphoma (DHL) and double-expressor (DEL) cell lines that were screened in current study and their basal levels of MYC and BCL2 expression. Cells were harvested while in exponential growth phases and Western blot analysis was performed with whole cell lysate.

CG-806 Inhibits Proliferation and Induces Apoptosis in Double-Hit and Double-Expressor Lymphoma Cells and Enhances Venetoclax Apoptotic Effect

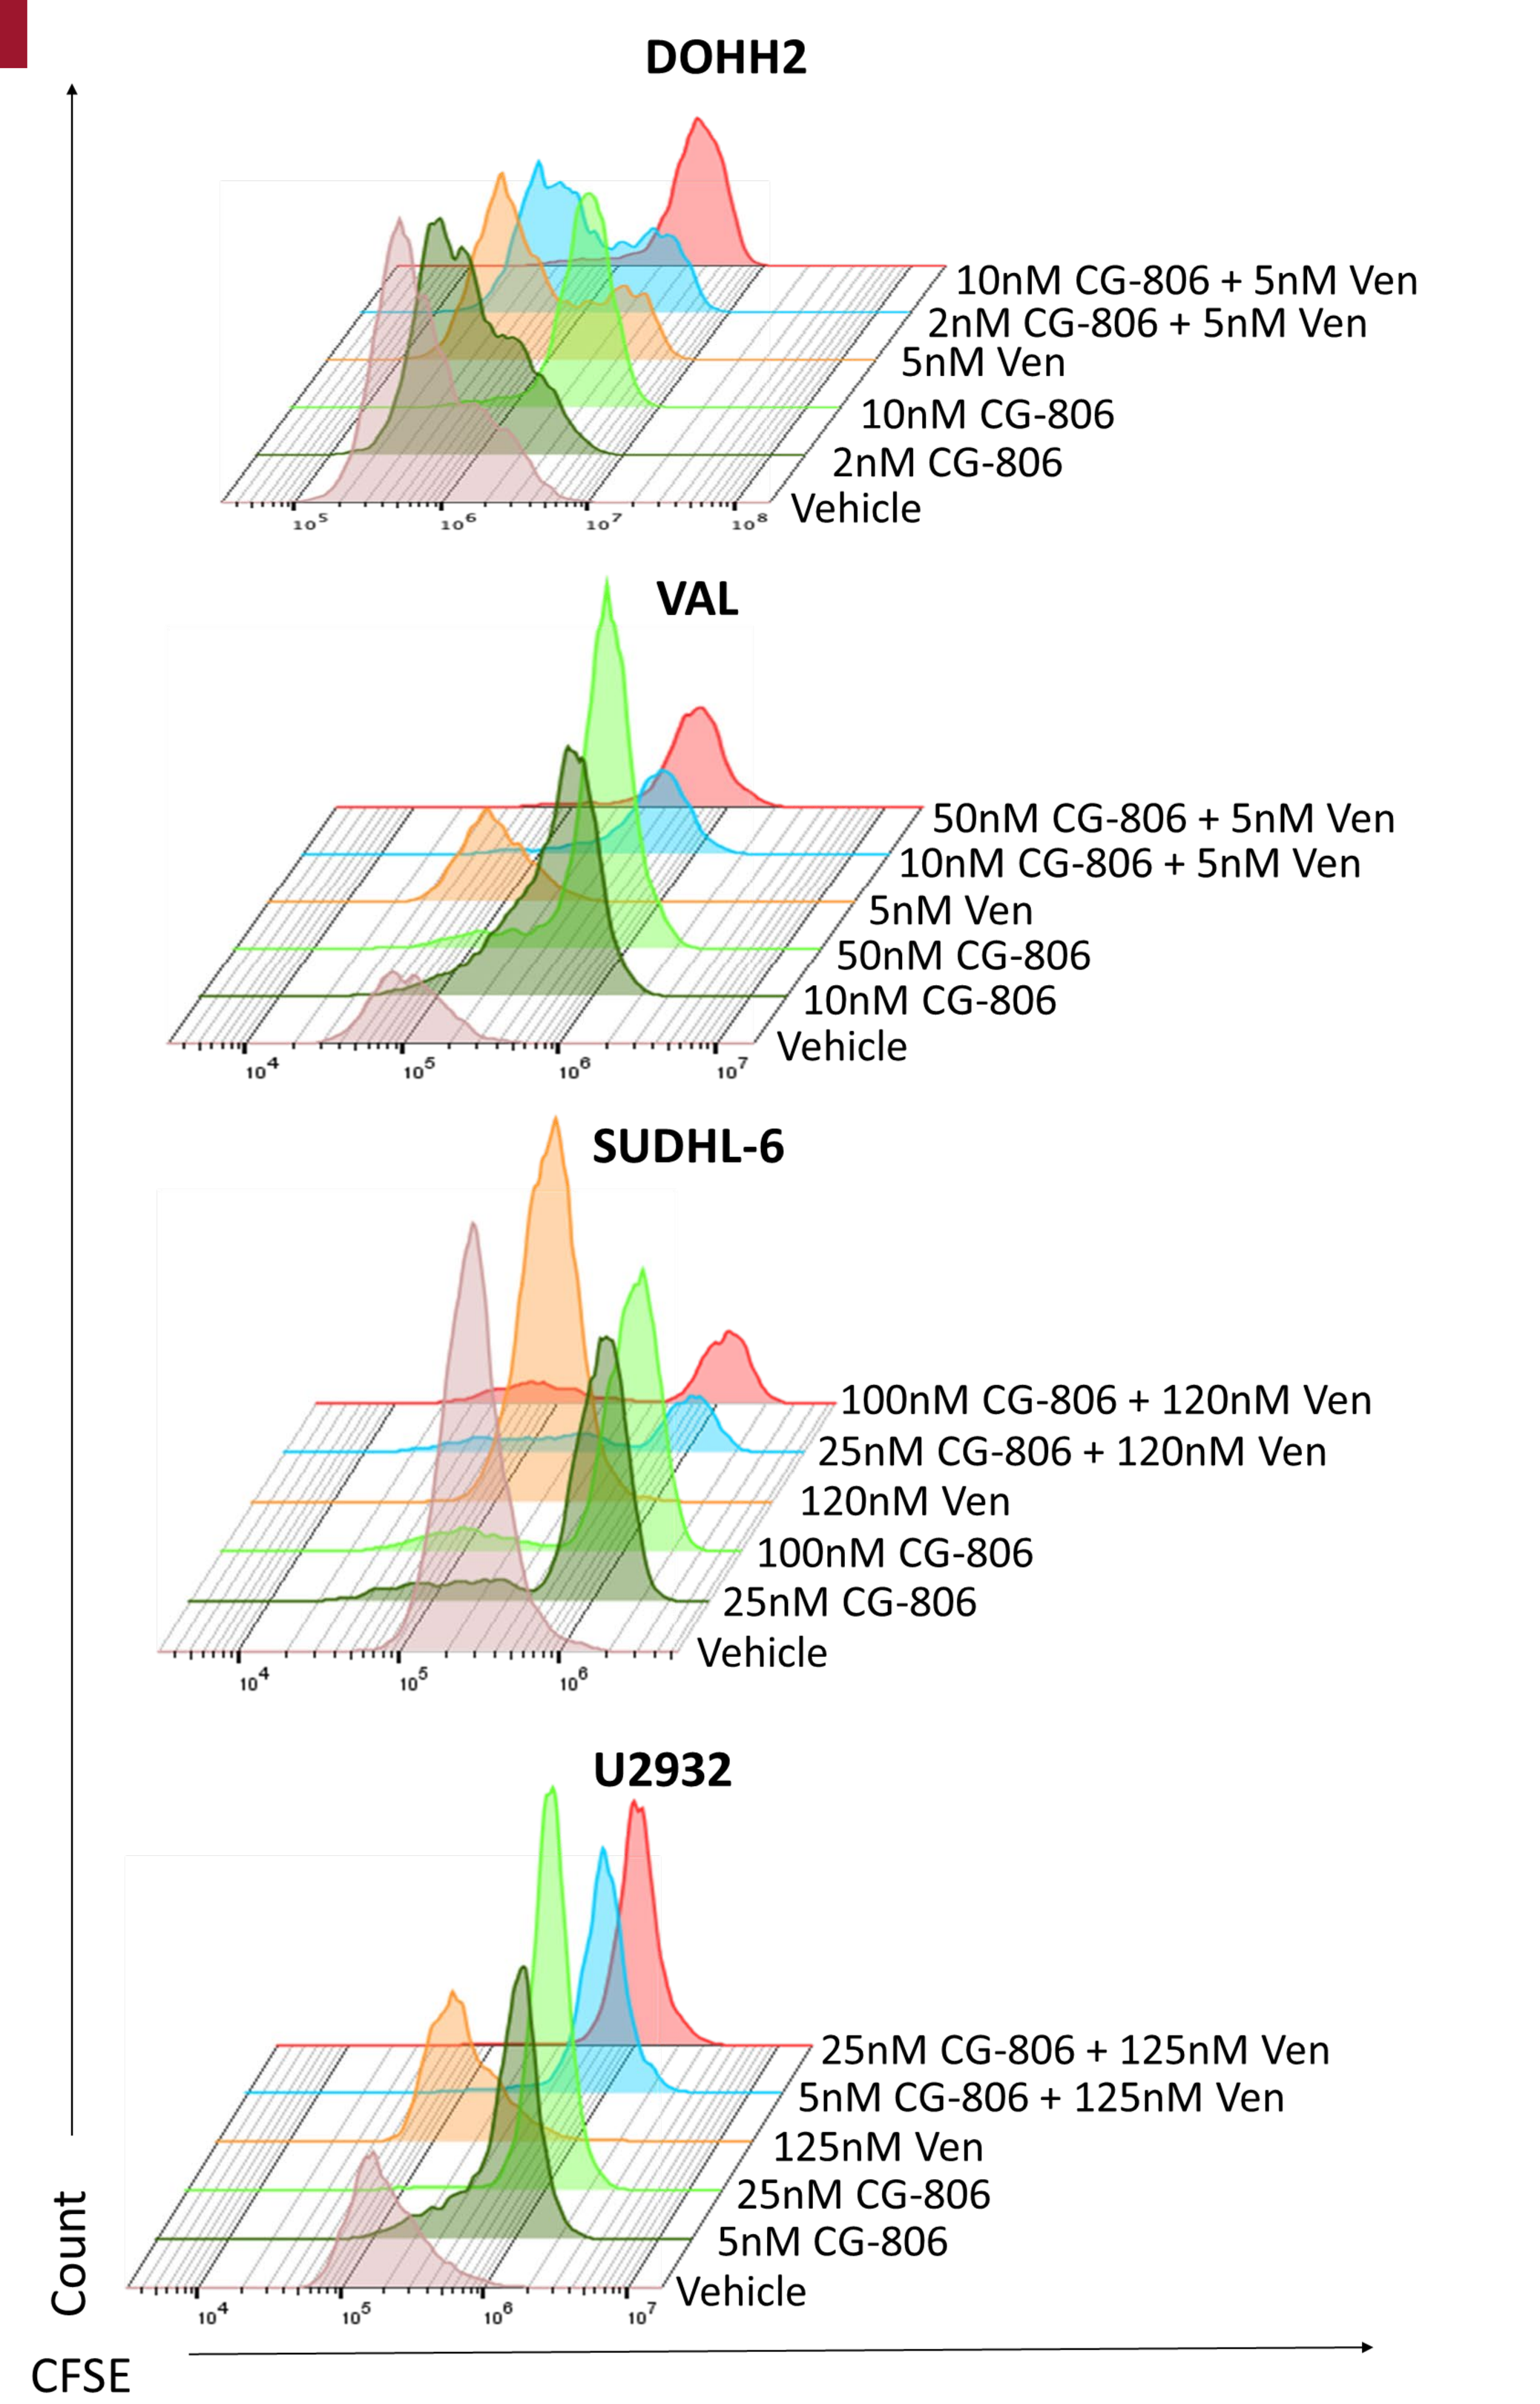
DOHH2
10nM CG-806 + 5nM Ven
2nM CG-806 + 5nM Ven
5nM Ven
10nM CG-806
2nM CG-806
Vehicle
VAL
50nM CG-806 + 5nM Ven
10nM CG-806 + 5nM Ven
5nM Ven
50nM CG-806
10nM CG-806
Vehicle
SUDHL-6
100nM CG-806 + 120nM Ven
25nM CG-806 + 120nM Ven
120nM Ven
100nM CG-806
25nM CG-806
Vehicle
U2932
25nM CG-806 + 125nM Ven
5nM CG-806 + 125nM Ven
125nM Ven
25nM CG-806
5nM CG-806
Vehicle
Count
CFSE

CG-806, but not Venetoclax, inhibits proliferation of DHL/DEL cells. Cells were stained with CFSE and treated with or without CG-806 alone or in combination with venetoclax for 72 h and subjected to cell proliferation analysis by flow cytometry.

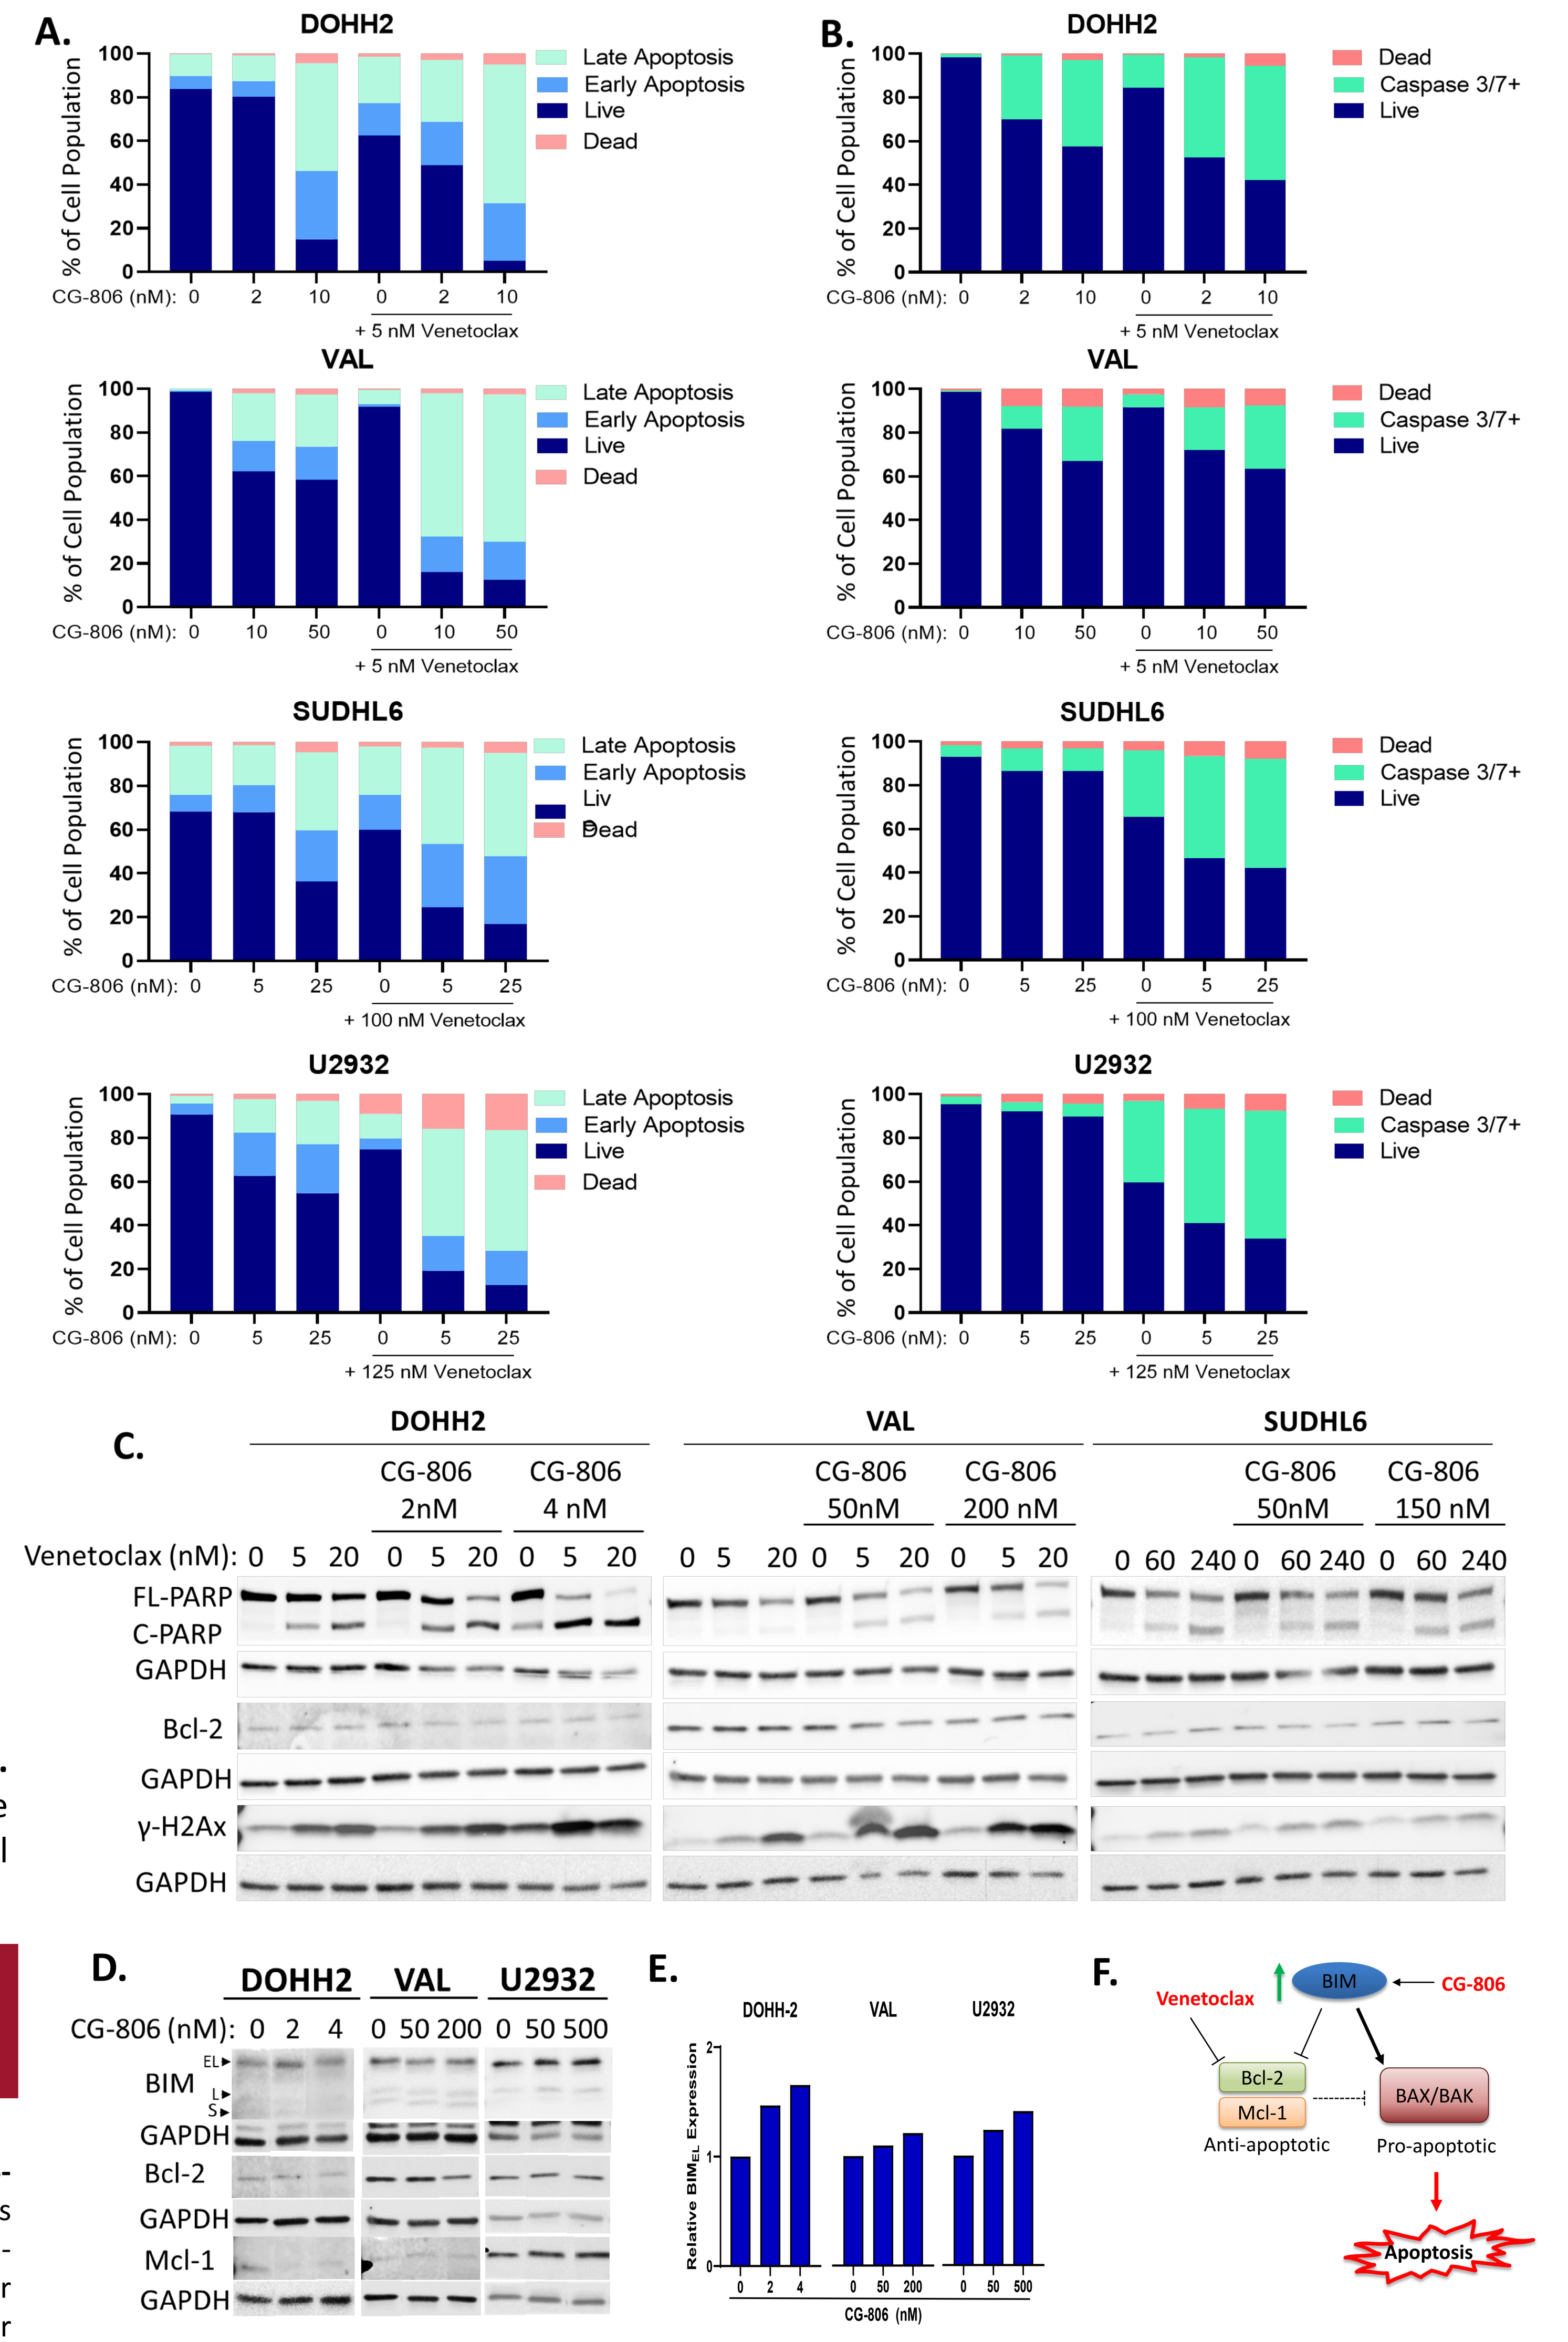
A.
B.
DOHH2
VAL
SUDHL6
U2932
Late Apoptosis
Early Apoptosis
Live
Dead
Caspase 3/7+
% of Cell Population
CG-806 (nM):
+ 5 nM Venetoclax
+ 100 nM Venetoclax
+ 125 nM Venetoclax
C.
CG-806 2nM
CG-806 4 nM
CG-806 50nM
CG-806 200 nM
CG-806 150 nM
Venetoclax (nM):
FL-PARP
C-PARP
GAPDH
Bcl-2
γ-H2Ax
D.
CG-806 (nM):
BIM
Mcl-1
E.
DOHH-2
Relative BIM_{EL} Expression
CG-806 (nM)
F.
Venetoclax
BIM
CG-806
Bcl-2
Mcl-1
Anti-apoptotic
BAX/BAK
Pro-apoptotic
Apoptosis

CG-806 induces apoptosis in DHL/DEL cells and enhances apoptotic effect of venetoclax. Cells were treated with or without CG-806 alone or combined with venetoclax and then subjected to annexin V / PI staining (Fig A. 72 h treatment) and caspase 3/7 activity flow cytometry assay (Fig B. 24 h treatment) and immunoblotting (Fig C and Fig D. 24 h treatment; Fig E. Quantification of BIM-EL isoform in Fig D). Schematic model of CG-806 increasing BIM in DHL/DEL to enhance venetoclax apoptotic effect (Fig F).

CG-806 Suppresses BCR Signaling and Compensatory Survival Pathways in Double-Hit and Double-Expressor Lymphoma Cells

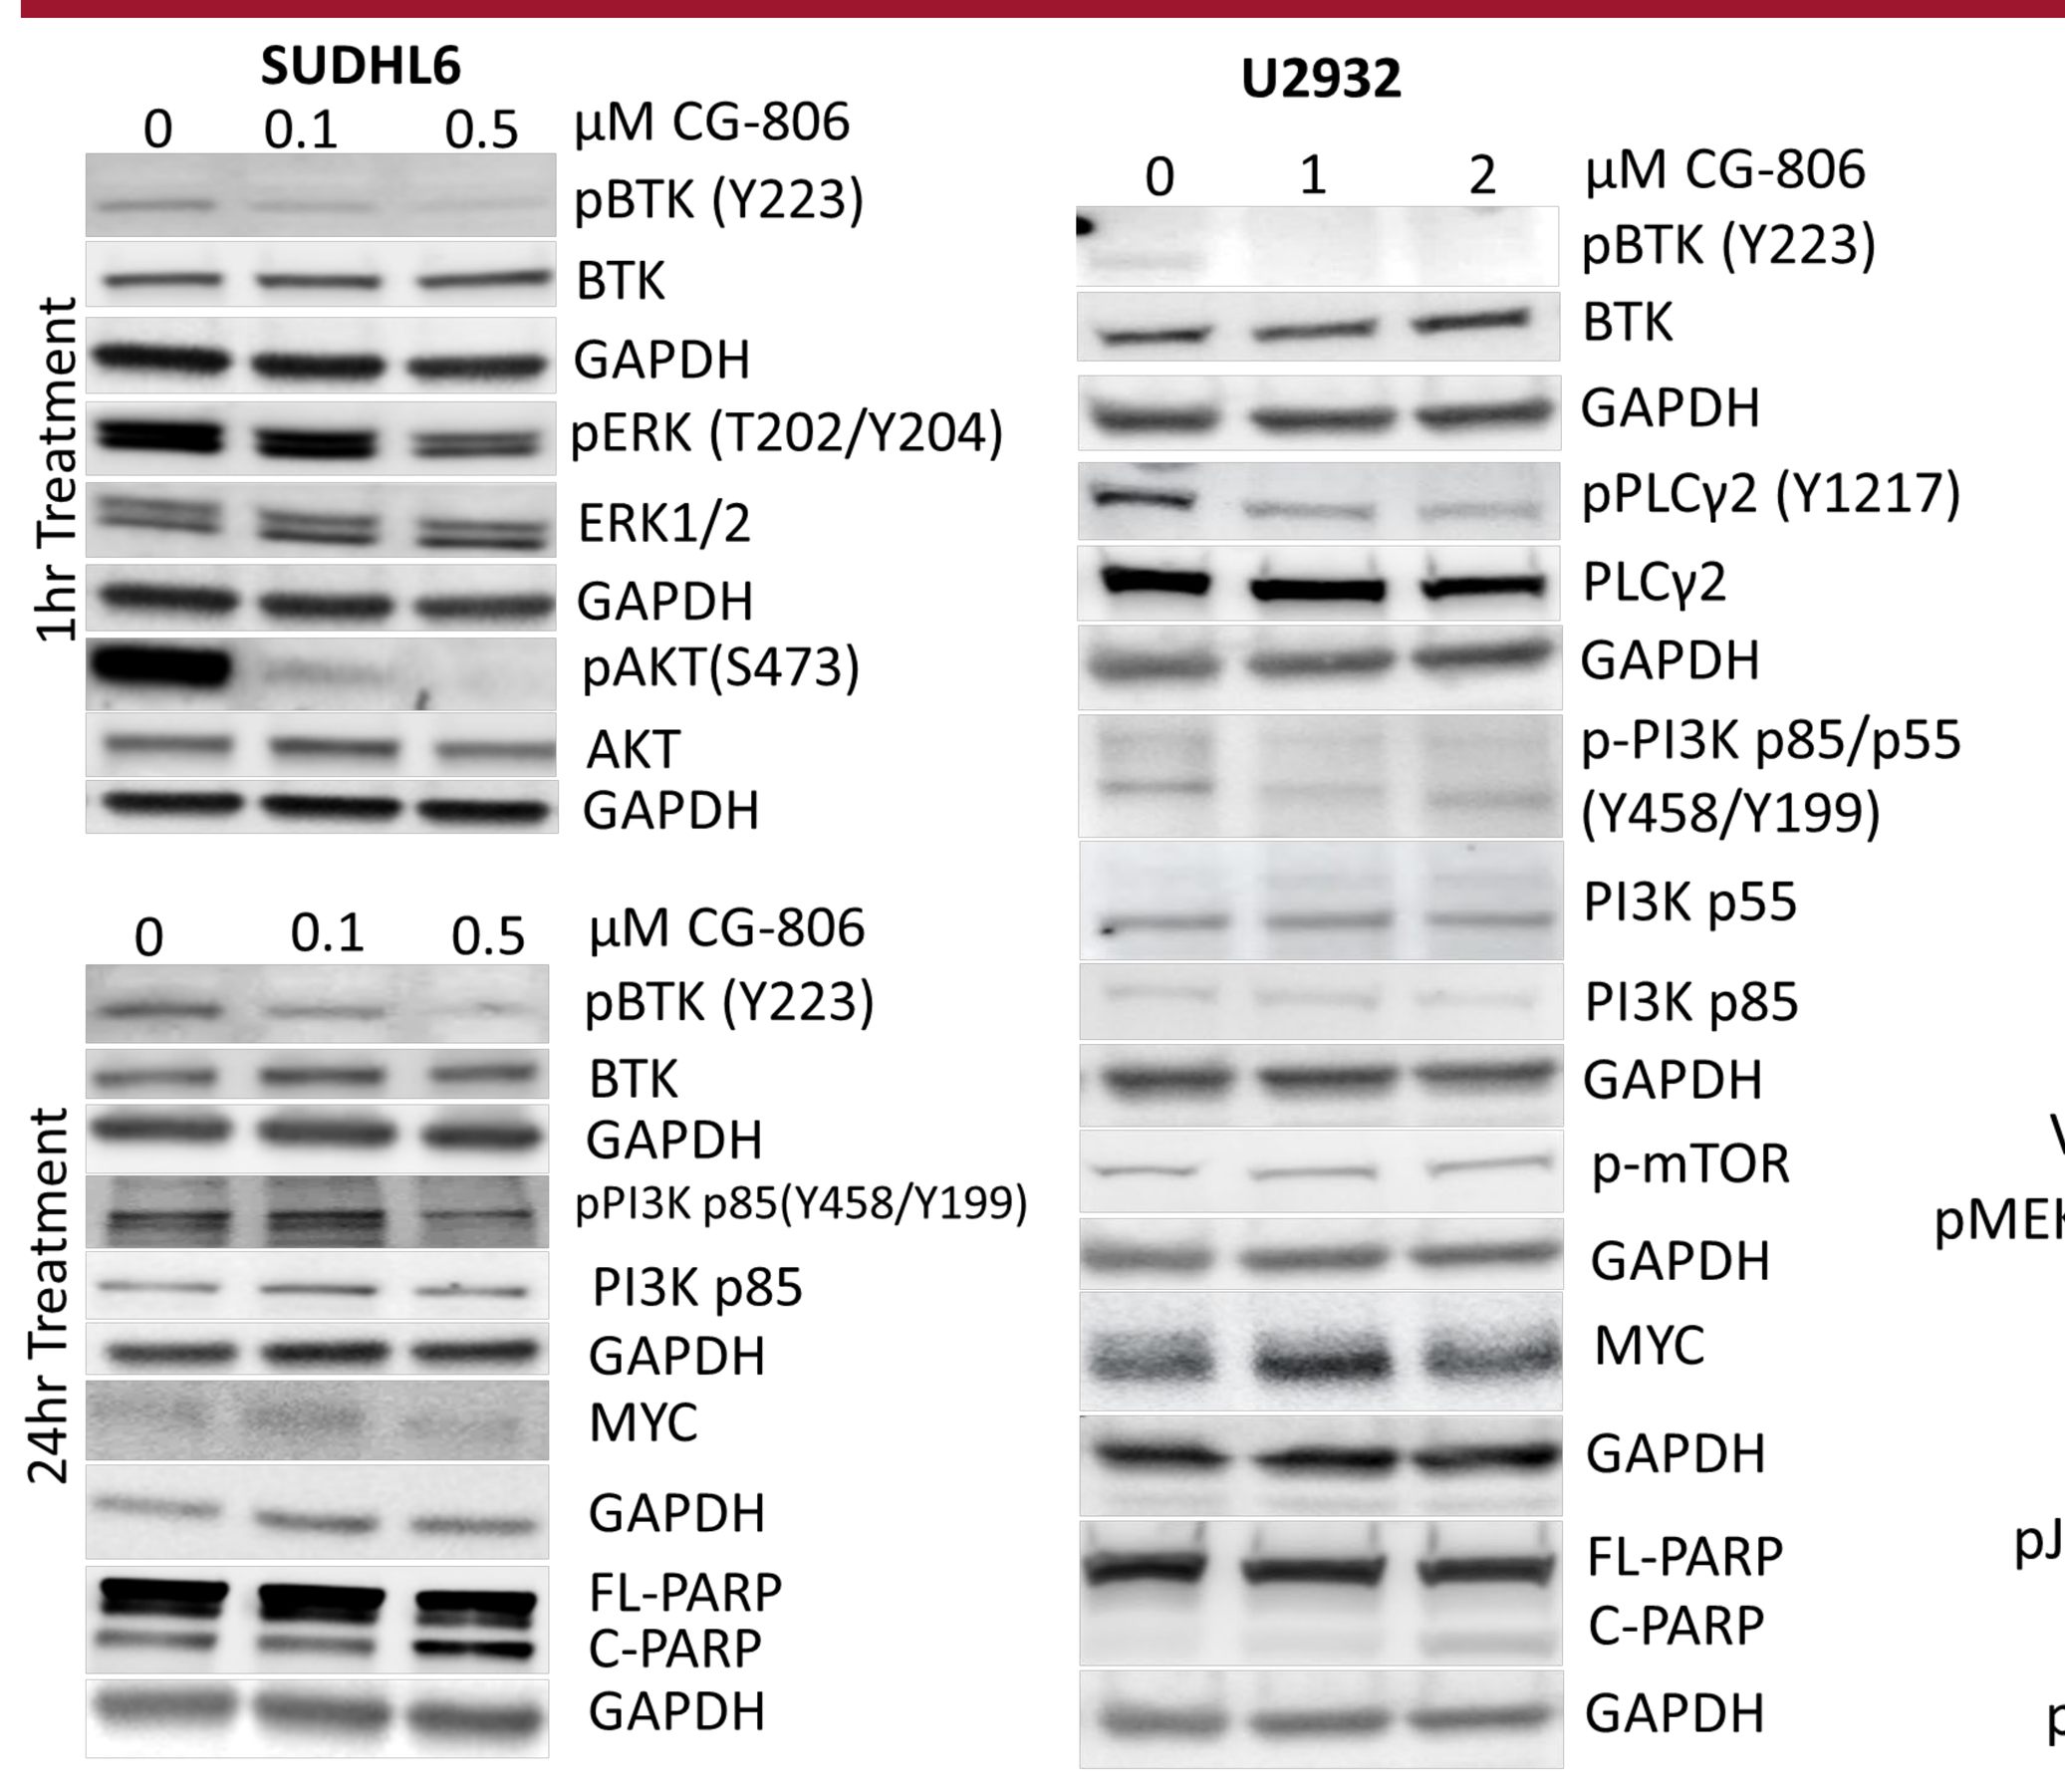
SUDHL6
U2932
1hr Treatment
24hr Treatment
µM CG-806
pBTK (Y223)
BTK
GAPDH
pERK (T202/Y204)
ERK1/2
pAKT(S473)
AKT
pPI3K p85(Y458/Y199)
PI3K p85
MYC
FL-PARP
C-PARP
pPLCγ2 (Y1217)
PLCγ2
p-PI3K p85/p55 (Y458/Y199)
PI3K p55
p-mTOR

CG-806 inhibits BCR and PI3K/AKT pathways, as demonstrated by reducing phosphorylated BTK, $PLC_{\gamma}2$, ERK, PI3K and AKT, and decreasing expression level of MYC. Cells were treated with or without CG-806 at indicated concentrations for indicated time and then subjected to immunoblotting.

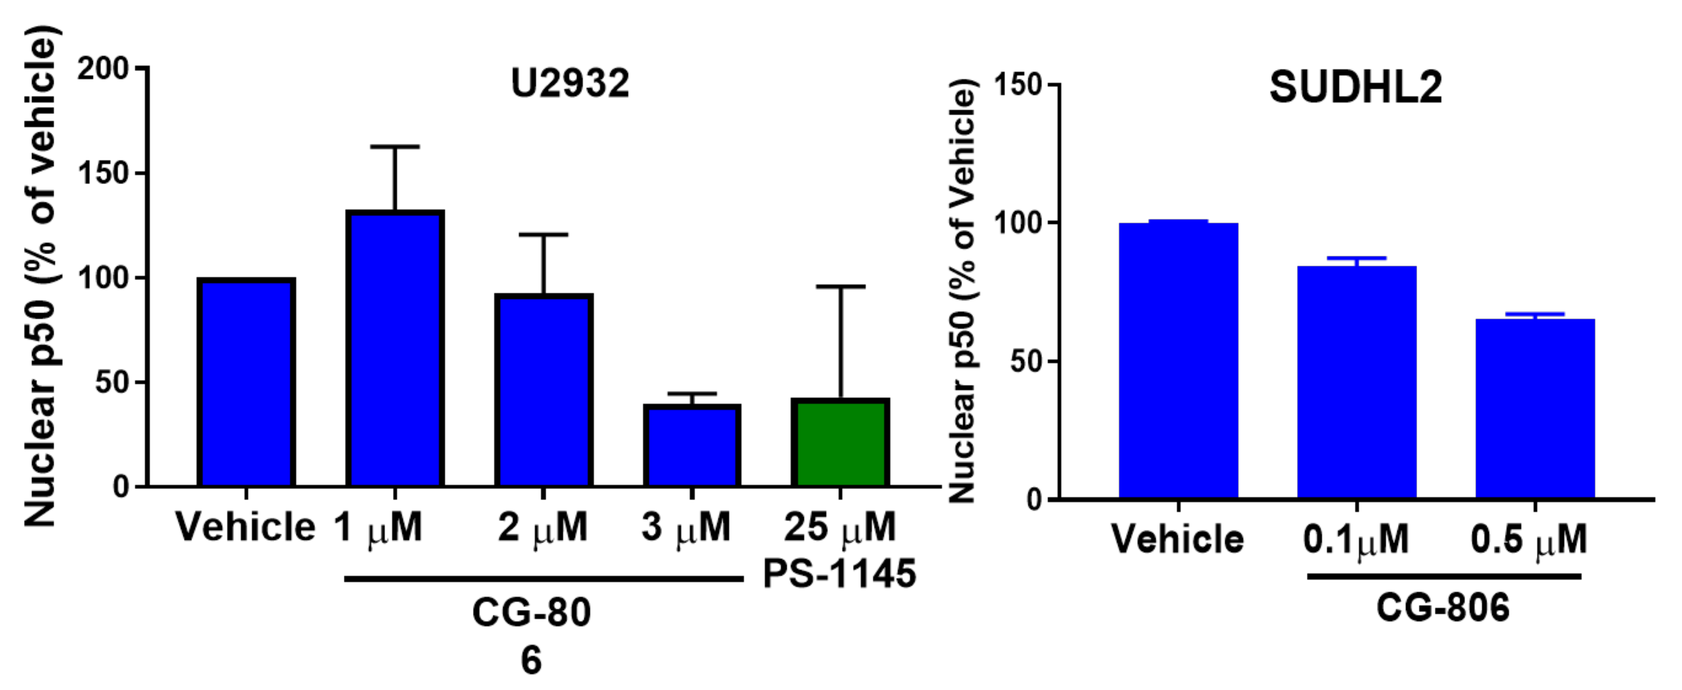
U2932
SUDHL2
Nuclear p50 (% of vehicle)
Vehicle
CG-806
PS-1145

Concentration-dependent decrease in NFκB activity after 6-hour CG-806 Treatment. Cells were treated with or without CG-806 at indicated concentrations for 6 h and then subjected to nuclear extract and NFκB p50 transcription factor activity assay. PS-1145, a specific IKK inhibitor, was used as a positive control.

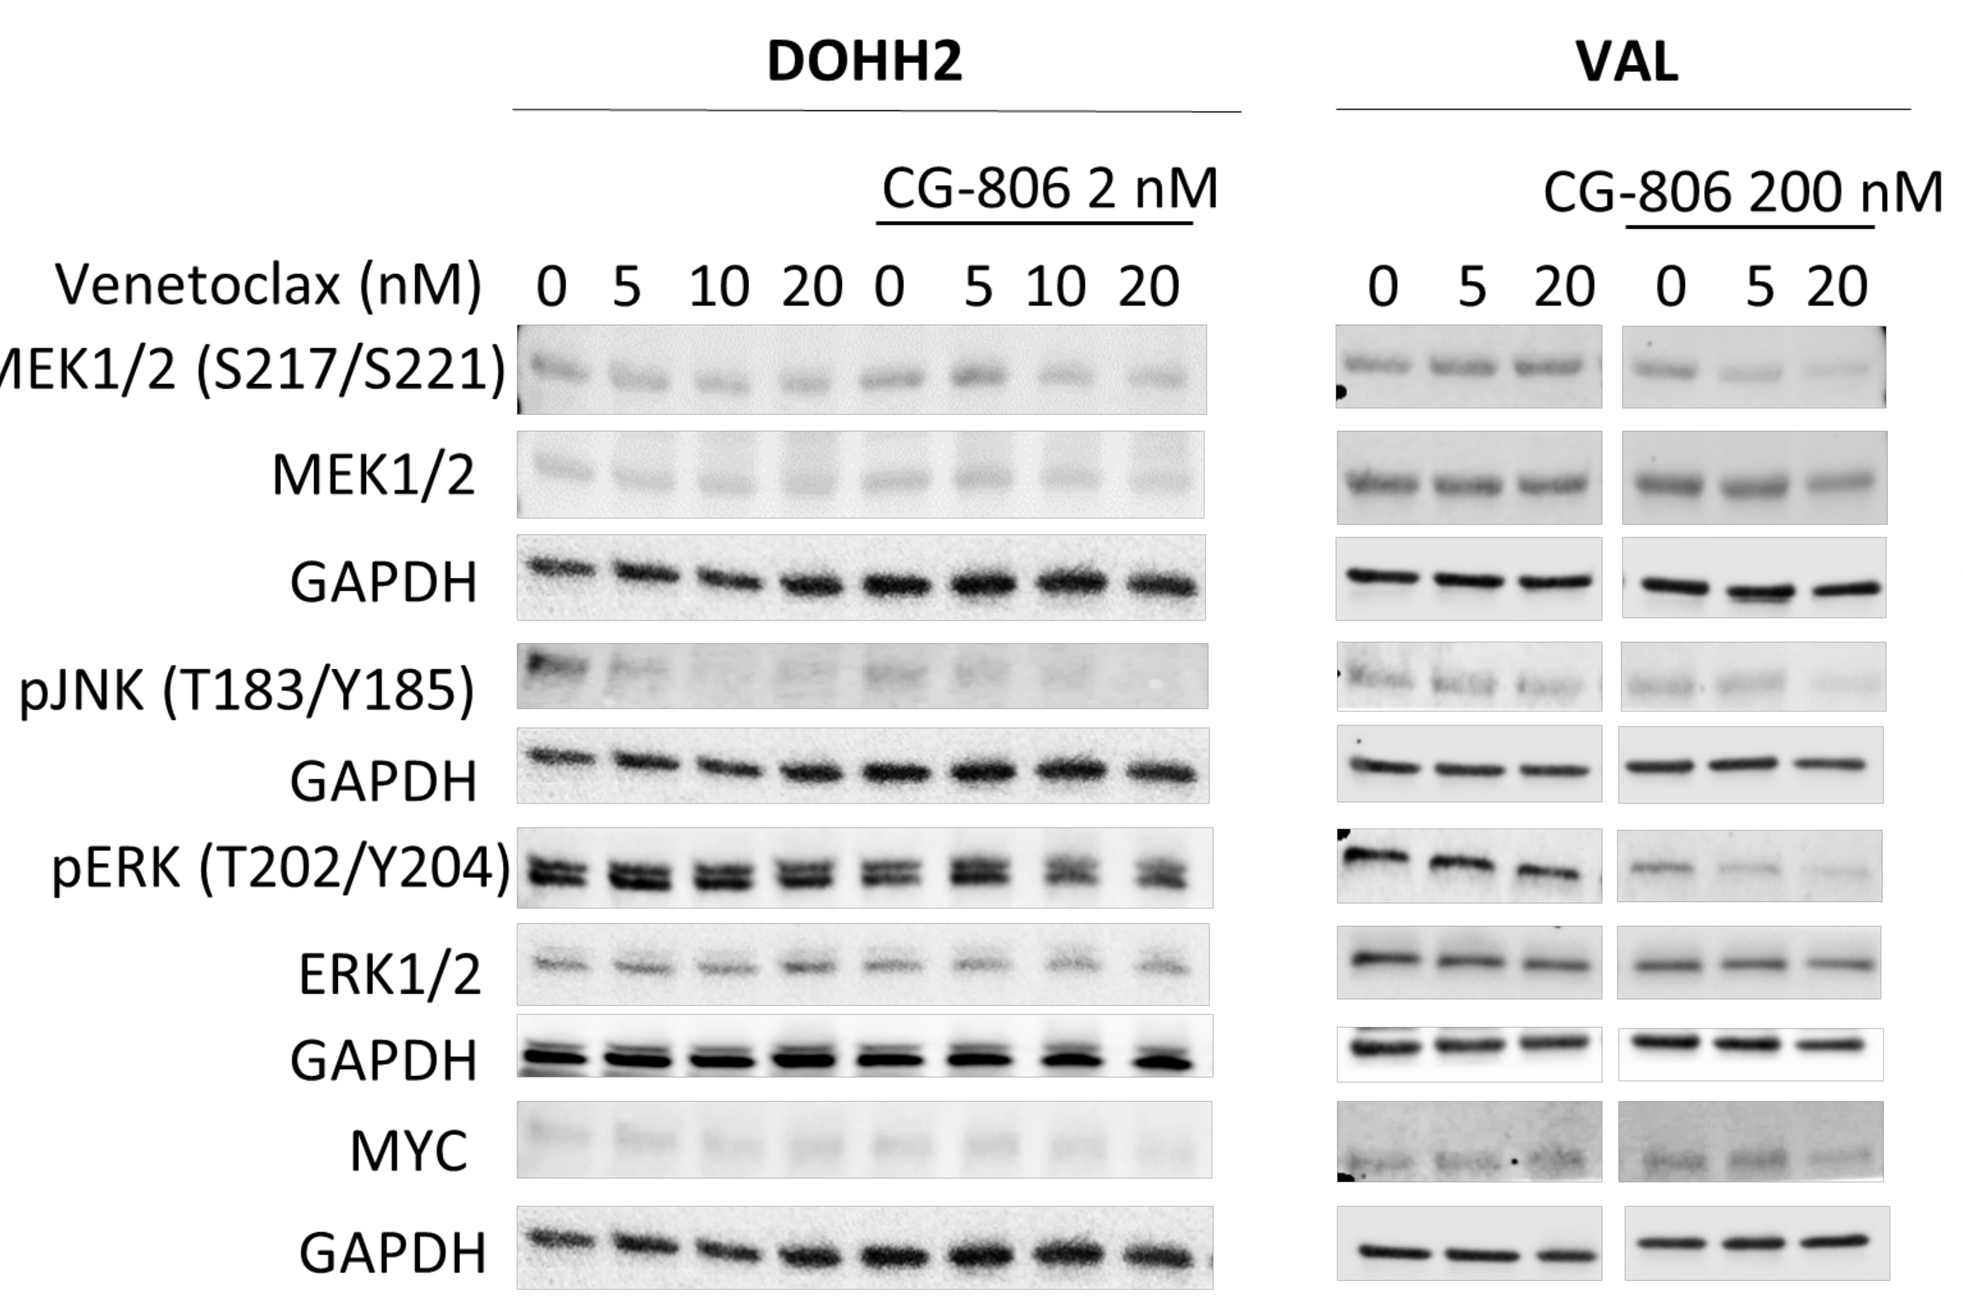
DOHH2
VAL
CG-806 2 nM
CG-806 200 nM
Venetoclax (nM)
pMEK1/2 (S217/S221)
MEK1/2
GAPDH
pJNK (T183/Y185)
pERK (T202/Y204)
ERK1/2
MYC

CG-806 inhibits MAPK signaling by reducing phosphorylated MEK1/2, JNK and ERK. Cells were treated with or without CG-806 alone or in combination with venetoclax at indicated concentrations for 24 h and then subjected to Western blot analysis using whole cell lysates.

Conclusions

- **CG-806 potently kills double-hit and double-expressor lymphoma cells and synergizes with venetoclax to inhibit cell proliferation and induce apoptosis.**
- **CG-806 inhibits driver BCR pathway and PI3K/AKT, NFκB and MAPK-mediated rescue pathways to kill aggressive B-cell lymphomas.**
- **CG-806 is currently being evaluated in a Phase 1 a/b trial in patients with CLL and relapsed or refractory B-cell malignancies (NCT03893682).**
- **The current study provides additional mechanistic evidences to support clinical development of CG-806 as a single agent or in combination with venetoclax in patients with aggressive B-cell lymphomas harboring unfavorable BCL2/MYC/BCL6 translocations and / or overexpression.**