Evaluation of Antifungal Activity of SCY-078 in Combination with Other Antifungals Against *Aspergillus* Strains

Lisa Long, BA 1; Emily L. Larkin, BA 1; Katyna Borroto-Esoda, PhD 2; Steve Wring, PhD 2; David Angulo, MD 2; and Mahmoud A. Ghannoum, PhD 1
1.Center for Medical Mycology, Case Western Reserve University and University Hospitals Cleveland Medical Center, Cleveland, OH; 2. Scynexis Inc., NJ, USA

27th ECCMID
Vienna, Austria
22 – 25 April 2017

INTRODUCTION & PURPOSE

Invasive aspergillosis (IA) is a fungal infection associated with high mortality, affecting vulnerable populations such as patients with hematologic malignancies and hematopoietic cell transplant recipients. The mortality remains high (>20%) in spite of available therapies including Voriconazole, Isavuconazole, Amphotericin B and the Echinocandins. Exploring new treatment paradigms for IA including combination therapy is needed to improve outcomes.

SCY-078 is an oral and intravenous semi-synthetic triterpenoid antifungal glucan synthase inhibitor, currently in development for the treatment of invasive and mucocutaneous fungal diseases. It has a broad-spectrum of activity against both *Aspergillus* and *Candida* species.

The objective of this study was to determine whether the combination of SCY-078 with Amphotericin B, Isavuconazole or Voriconazole would increase their antifungal activity *in vitro* against *Aspergillus fumigatus.*

MATERIALS & METHODS

- Six strains of *A. fumigatus* were tested: 4 wild-type (WT) and 2 with elevated azole MICs (Azole-R) – one of which has a CYP51 mutation at F46Y (strain 28500)
- MIC determinations (at 48h) of the individual antifungals alone and when in combination were determined following a checkerboard design[2]
- The effect of combination testing was reported according to an Fractional Inhibitory Concentration Index (FICI),, which assigns a numerical value (formula below) to the interaction of the two compounds
- FICI interpretation:
 - Synergistic FICI ≤ 0.5
 - Additive FICI > 0.5 but ≤ 4.0
 - Antagonistic FICI > 4.0

RESULTS

Table 1. MIC values (µg/mL) alone & in combination for SCY-078 with other antifungal agents against *A. fumigatus* (test performed in duplicate, representative value displayed)

Strain	SCY-078 with Isavuconazole (ISA)						SCY-078 with Voriconazole (VRC)						SCY-078 with Amphotericin B (AmB)					
	MIC Alone		MIC Combo		FICI	Interpretation*	MIC Alone		MIC Combo		FICI	Interpretation*	MIC Alone		MIC Combo		FICI	Interpretation*
	SCY-078	ISA	SCY-078	ISA	SCY-078 + ISA		SCY-078	VRC	SCY-078	VRC	SCY-078 + VRC		SCY-078	AmB	SCY-078	AmB	SCY-078 + AmB	
WT 20438	4	1	0.016	0.5	0.50	SY	4	1	0.125	0.25	0.27	SY	4	4	0.016	0.5	0.13	SY
WT 28378	4	1	0.125	0.25	0.28	SY	4	0.25	0.5	0.16	0.19	SY	4	2	0.016	0.5	0.25	SY
WT 28382	4	1	0.063	0.25	0.27	SY	8	0.5	0.5	0.125	0.31	SY	4	4	0.016	1	0.25	SY
WT 28401	4	1	0.25	0.25	0.31	SY	8	2	0.25	0.5	0.28	SY	4	4	0.016	1	0.25	SY
Azole-R 28383	4	>8	0.063	>8	1.02	AD	8	>16	0.031	>16	1.00	AD	4	2	0.125	2	1.03	AD
Azole-R 28500	4	>8	0.125	>8	1.03	AD	4	>16	1	>16	1.25	AD	4	4	0.016	1	0.25	SY

* SY= synergistic, AD = additive, AN = antagonistic

CONCLUSIONS

- SCY-078 in combination with Voriconazole, Isavuconazole and Amphotericin B demonstrates synergistic activity against the majority of *A. fumigatus* isolates tested
- For azole-resistant strains, additive effect was observed
- SCY-078 showed no *in vitro* antagonism with any of the drugs tested
- Results warrant subsequent evaluations of SCY-078 in combination with mold-active antifungal agents for the treatment of IA

References: 1. **Johan Maertens et al.** 2015. Combination antifungal therapy of Invasive Aspergillosis. Annals of Internal Medicine **162:**81-89. 2. **Mukherjee, P. K., D. J. Sheehan, C. A. Hitchcock, and M. A. Ghannoum.** 2005. Combination treatment of invasive fungal infections. Clin Microbiol Rev **18:**163-94.

For additional information contact Mahmoud A. Ghannoum, PhD at mag3@case.edu or Scynexis at info@scynexis.com.

EV0123