A Recombinant Human Antibody to EphA3 with Pro-Apoptotic and Enhanced ADCC Activity Shows Selective Cytotoxicity against Myeloid Leukemia Cells and CD123-Positive Leukemic Stem Cells

Varghese Palath, PhD[1], Rohini Vekhande[1], Andreia Lee[1], Jason Williams[1], Ling Zhang, MD[2], Alan F List, MD[3], Andrew Boyd, MD, PhD[4], Martin Lackmann, PhD[5], Andrew M Scott, PhD[6], Daniela Cilloni, MD, PhD[7], Geoffrey T. Yarranton, PhD[1] and Christopher Bebbington, DPhil[1]

[1]KaloBios Pharmaceuticals, Inc., South San Francisco, CA; [2]Department of Pathology & Lab, H. Lee Moffitt Cancer Center, Tampa, FL; [3]H. Lee Moffitt Cancer Center, Tampa, FL; [4]Queensland Institute for Medical Research, Brisbane, Queensland, Australia; [5]Monash University, Clayton, Victoria, Australia; [6]Ludwig Institute for Cancer Research, Melbourne, Australia; [7]Department of Clinical and Biological Sciences, University of Turin, Orbassano, Turin, Italy

ABSTRACT

Members of the Eph family of receptor tyrosine kinases play important roles in embryonic development but have restricted tissue distribution and activity in adult tissues. The EphA3 receptor is an oncofetal antigen expressed at high levels on the surface of several solid tumor types and certain leukemias.

KaloBios is in pre-clinical development with KB004, a high-affinity recombinant human antibody, derived from a monoclonal anti-EphA3 antibody by Antibody Humaneering™ Technology. KB004 binds EphA3 and stimulates apoptosis in primary cells from myeloid leukemia patients. The cell-surface expression of EphA3 was analyzed by flow cytometry on primary cells from chronic myeloid leukemia (CML) [n=10], acute myeloid leukemia (AML) of various sub-types [n=37], and myelodysplastic syndromes (MDS) [n=16]. EphA3 surface expression was detected in at least 50% of patient samples from each of the diseases analyzed. CD123-positive leukemia stem cells ($CD34^+$ $CD38^-$ $CD123^+$) also displayed surface expression of EphA3 whereas normal bone marrow $CD34^+$ stem/ progenitor cells lacked detectable EphA3. KB004 was shown to stimulate apoptosis in $EphA3^+$ primary leukemia cells, including $CD123^+$ leukemic stem cells with no activity in EphA3-negative specimens, including normal $CD34^+$ bone marrow cells. Cross-linking of the antibody was not required for induction of apoptosis. KB004 also inhibited myeloid leukemia colony formation (CFU-L) from primary AML samples in methylcellulose colony assays without affecting normal hematopoietic colony formation.

In addition to its direct pro-apoptotic effect, KB004 induced potent antibody-dependent cellular cytotoxicity (ADCC) activity against EphA3-positive cells, mediated by CD16-expressing effector cells. CD16-dependent ADCC activity was further enhanced by expression of the antibody in CHO cells that lack α1,6 fucosyl transferase, generating afucosylated antibody with higher affinity for CD16a.

The data support the development of an anti-EphA3 antibody for the treatment of hematologic malignancies in addition to solid tumors. In particular, the selective activity against leukemic stem cells supports a novel strategy for therapeutic targeting of leukemia-initiating cells that merits clinical investigation.

INTRODUCTION

EphA3 was originally identified as an oncofetal antigen on the pre-B-cell leukemic cell line (LK63) (Boyd *et al* 1992; Wicks *et al* 1992) using a monoclonal antibody IIIA4. The antibody has been shown to recognize the highly conserved N-terminal domain of EphA3 adjacent to the ligand binding site but does not interfere with binding of the natural ligand, ephrin-A5 (Smith *et al* 1994). KB004 is a high-affinity recombinant human IgG1 antibody, with the binding specificity of IIIA4, derived by Antibody Humaneering™ Technology. This technology replaces non-human V-segments with sequences from a human V-region library such that the V-regions of the resulting antibody have high sequence identity with human germ-line sequence and are expected to show low immunogenicity in humans.

The antibody is produced from a CHO cell line deficient in α1,6 fucosyl transferase to generate antibody lacking fucose in its carbohydrate structure. Non-fucosylated antibodies have been shown to have enhanced ADCC activity mediated by CD16a (Mori *et al.* 2007). Here we show the *in vitro* activity of KB004 against a panel of primary cell samples from myeloid leukemias, myelodysplastic syndrome (MDS) and other myeloproliferative neoplasms (MPN).

RESULTS

Figure 1. EphA3 is expressed on the cell surface in a proportion of primary samples from myeloid leukemias, myelodysplastic syndrome (MDS) and myeloproliferative neoplasms (MPN).

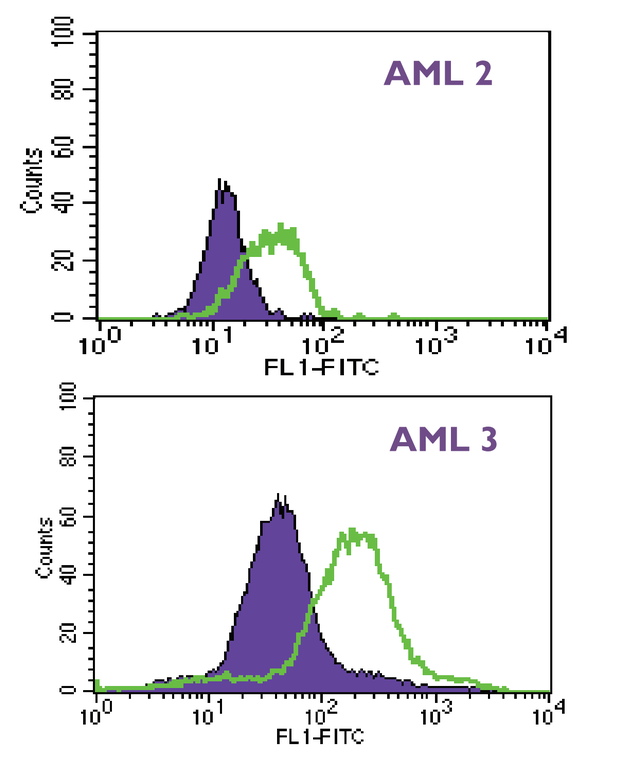
AML 2
AML 3
FL1-FITC

Tumor type	Number of samples tested	EphA3 positive samples	Samples positive for EphA3 (%)
AML	37	22	59
CML	10	5	50
MDS	16	7	44
MPN	6	3	50

Patient samples (bone marrow or PBMC) were stained with Humaneered anti-EphA3 antibody or isotype control IgG1 and FITC-conjugated anti-human IgG and analyzed by flow cytometry (10,000 to 50,000 events per sample). Histograms for two AML patients are shown. Samples were scored positive if at least 5% of cells showed higher fluorescence than the IgG1 control gate.

Figure 2. Anti-EphA3 antibody binds to AML bone marrow $CD34^+$ cells and cells with a leukemic stem cell phenotype ($CD34^+$ $CD38^-$ $CD123^+$).

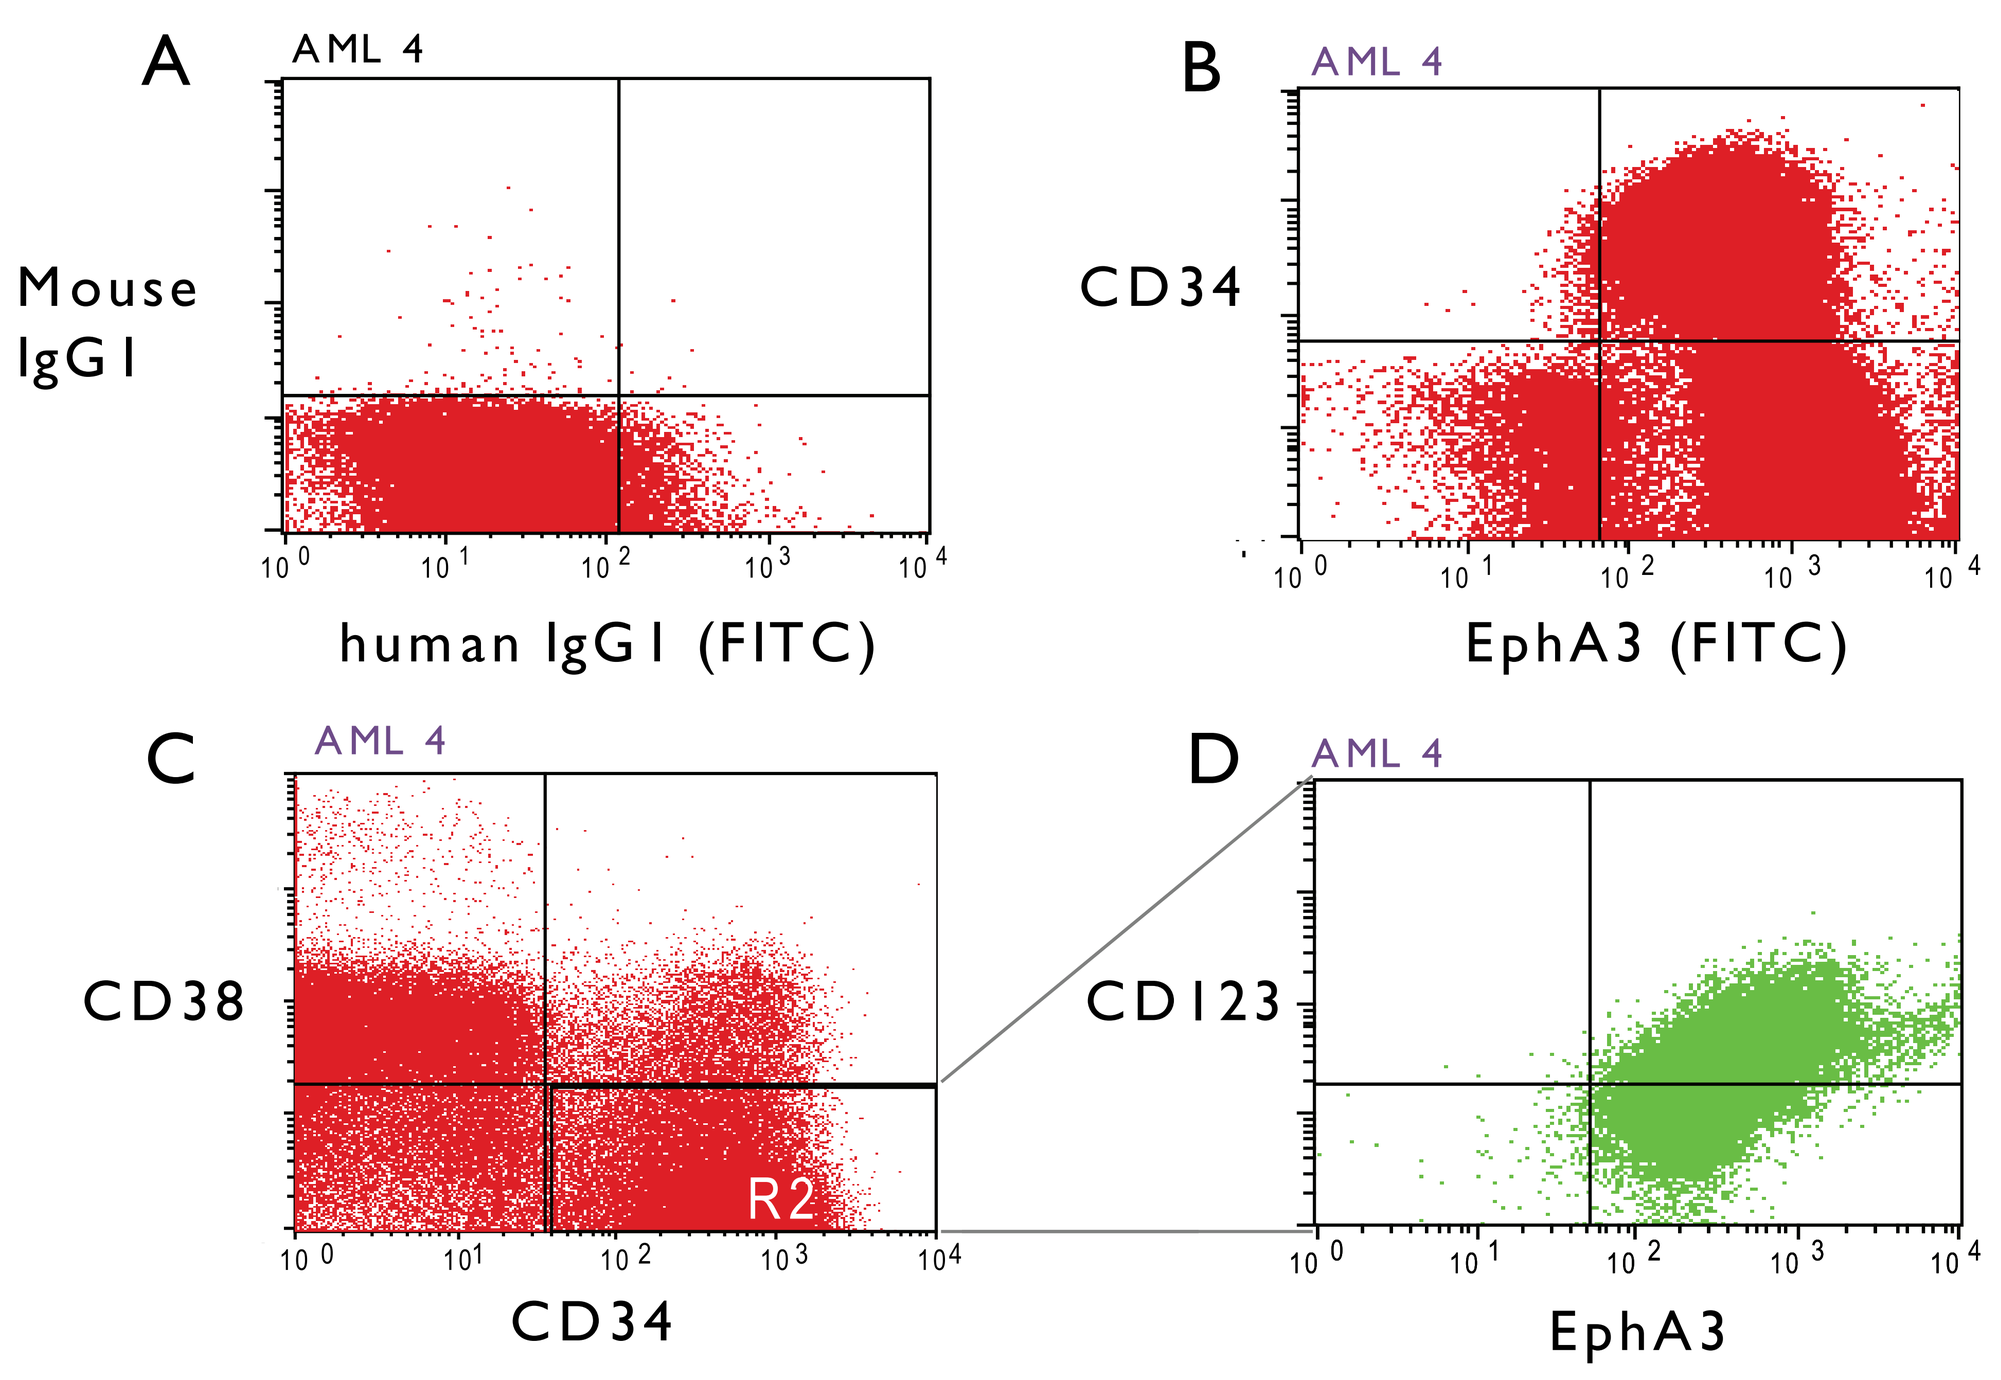
A
AML 4
Mouse IgG1
human IgG1 (FITC)
B
AML 4
CD34
EphA3 (FITC)
C
AML 4
CD38
R2
CD34
D
AML 4
CD123
EphA3

AML primary bone marrow cells were stained with: Humaneered anti-EphA3 antibody or IgG1 control and FITC-conjugated anti-human IgG; PE-conjugated anti-CD34; PEcy5-conjugated anti-CD38; and APC-conjugated anti-CD123 antibodies for flow cytometry analysis (50, 000 events per sample). (A) isotype control gating for CD34 analysis. (B) Sample stained with anti-EphA3 and anti-CD34. (C) Sample stained for CD34 and CD38 (R2 represents $CD34^+$ $CD38^-$ cells). (D) Identification of EphA3 and CD123 expression on $CD34^+$ $CD38^-$ cells (R2 gate).

Figure 3. EphA3 is not detectable on normal human bone marrow $CD34^+$ cells.

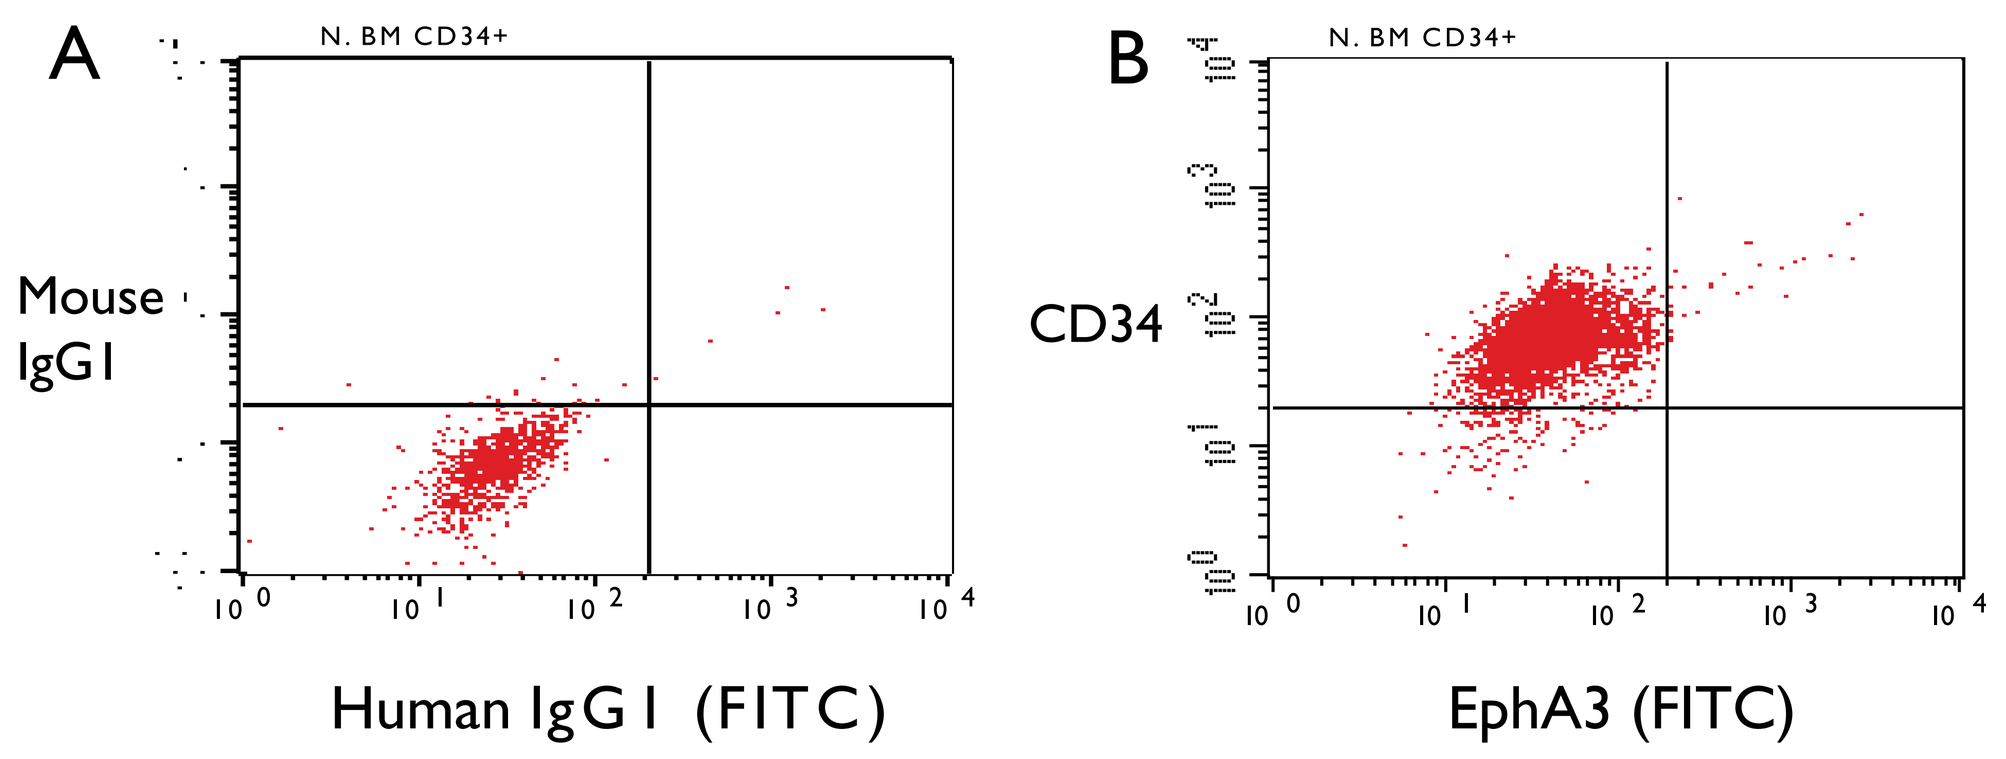
A
N. BM CD34+
Mouse IgG1
Human IgG1 (FITC)
B
N. BM CD34+
CD34
EphA3 (FITC)

Normal human bone marrow $CD34^+$ cells were stained with Humaneered anti-EphA3 antibody or control IgG1 and FITC-conjugated anti-human IgG and PE-conjugated anti-CD34 antibody and analyzed by flow cytometry (5000 events per sample). (A) isotype control gating for $CD34^+$ and EphA3 cell analysis. (B) staining with anti-EphA3 and anti-CD34.

Figure 4. Humaneered anti-EphA3 antibody induces apoptosis in $EphA3^+$ primary leukemia cells.

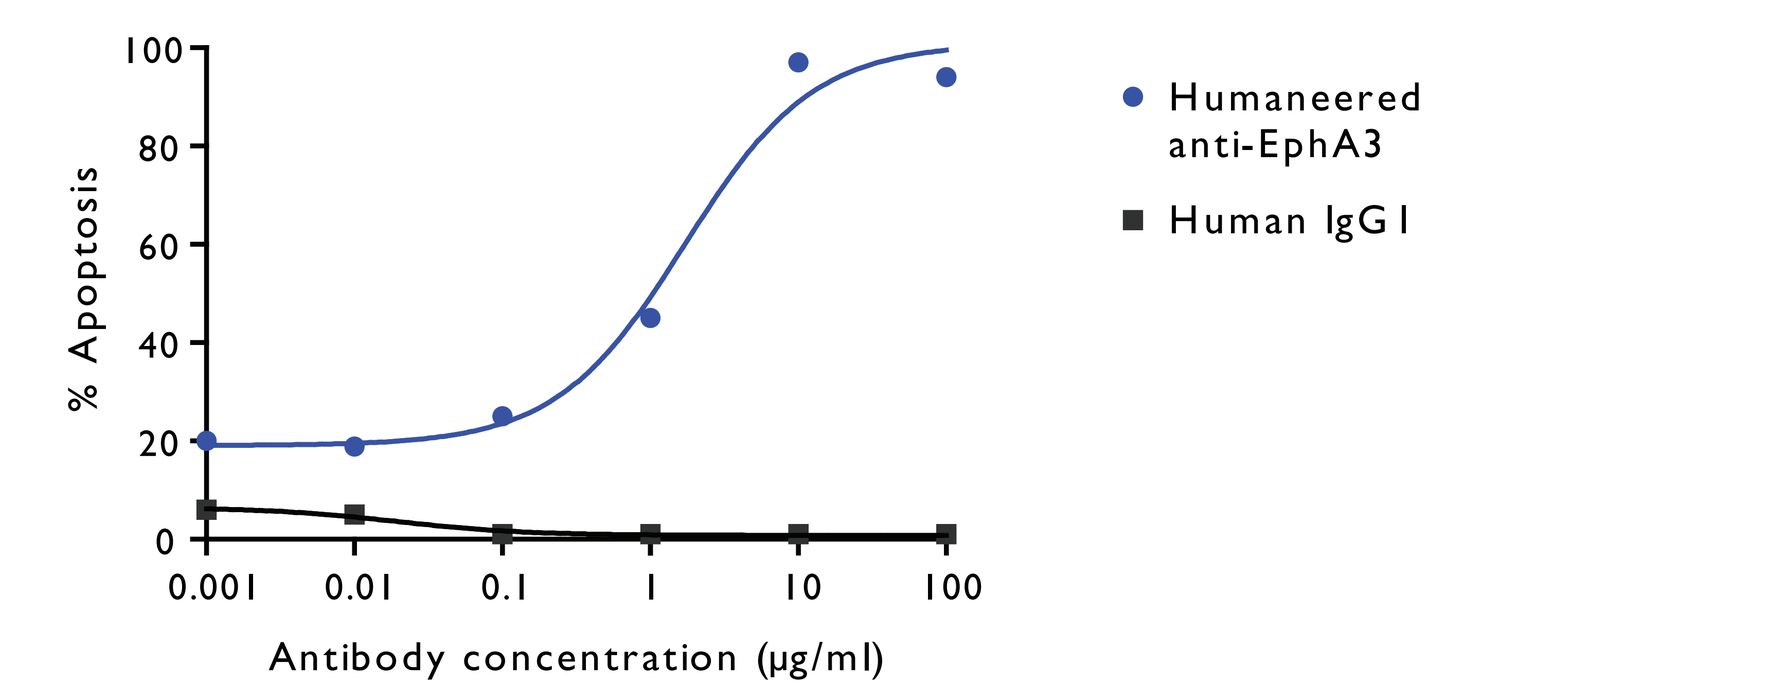
% Apoptosis
Antibody concentration (µg/ml)
Humaneered anti-EphA3
Human IgG1

Bone marrow cells from a CML patient (2x10^5 per well) were incubated with Humaneered anti-EphA3 antibody or IgG1 control antibody for 24 hours and stained with Annexin V-FITC and propidium iodide. Percent cells undergoing apoptosis (Annexin V-positive) are shown. The sample was 98% $EphA3^+$ by flow cytometry.

Figure 5. Humaneered anti-EphA3 Antibody induces ADCC mediated by CD16 (FcRIII). Non-fucosylated antibody (KB004) has enhanced ADCC activity.

A) CD16 dependent ADCC activity (CML PBMC target cells)

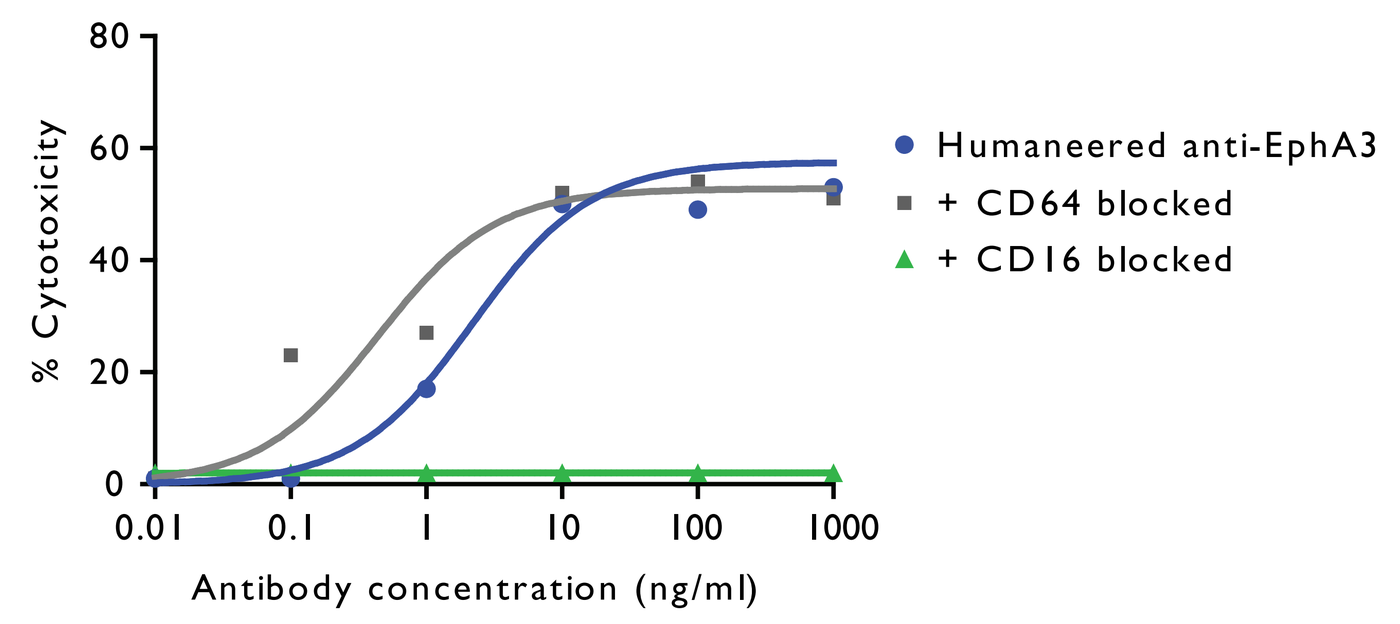
% Cytotoxicity
Antibody concentration (ng/ml)
Humaneered anti-EphA3
+ CD64 blocked
+ CD16 blocked

B) Enhanced ADCC activity by Anti-EphA3 lacking fucose (AML BM target cells)

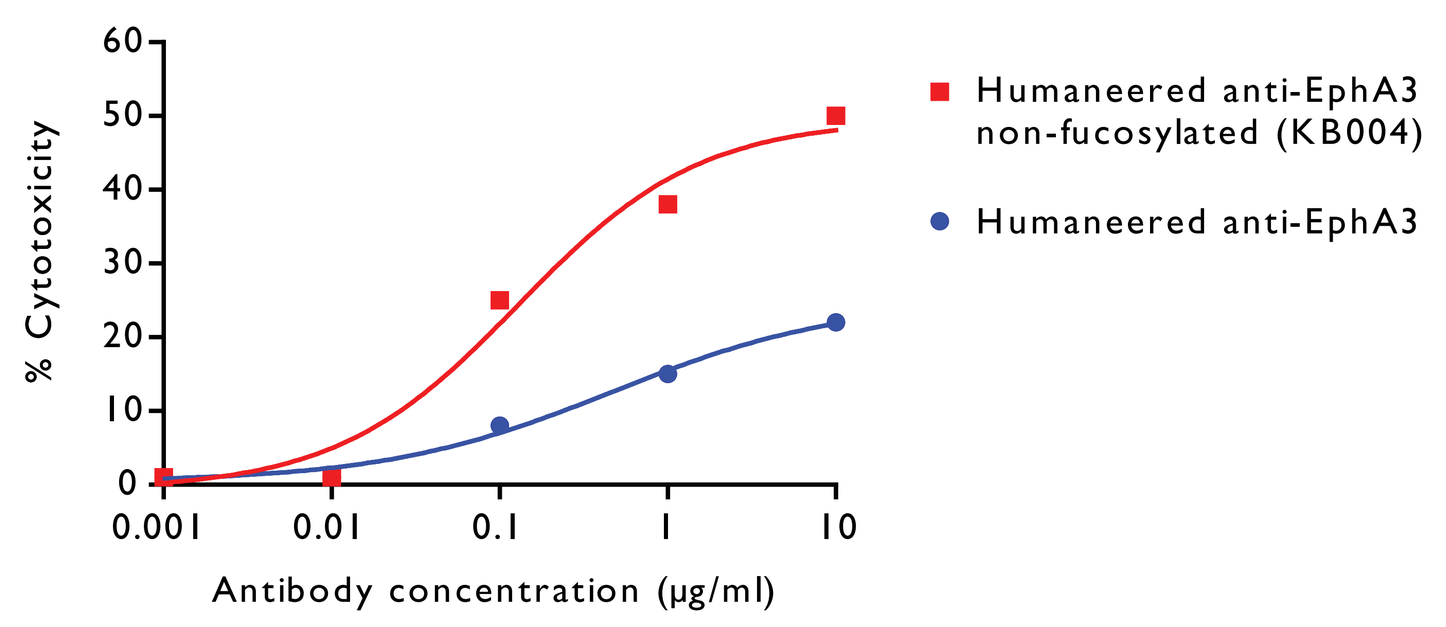
% Cytotoxicity
Antibody concentration (µg/ml)
Humaneered anti-EphA3 non-fucosylated (KB004)
Humaneered anti-EphA3

A) ADCC assay using CML PBMC as the target cells. 10^4 target cells were incubated with Humaneered Anti-EphA3 at different concentration for 16 hours with normal PBMC effector cells (CD16a genotype 158V/F) at an effector: target cell ratio of 100:1 in the presence or absence of neutralizing anti-CD16 (FcRIII) or anti-CD64 (FcRI) antibodies. Cytotoxicity was determined by LDH release. Target cells were 67% $EphA3^+$ by flow cytometry.

B) ADCC assay using AML bone marrow cells as the target. 10^4 AML bone marrow cells were incubated with Humaneered anti-EphA3 or non-fucosylated Humaneered anti-EphA3 antibody and normal PBMC effector cells (CD16a genotype 158F/F) at an effector: target ratio of 100:1 and LDH release was determined. Target cells were 41% $EphA3^+$ by flow cytometry.

Figure 6. KB004 antibody shows dose-dependent inhibition of leukemia colony formation in Methylcellulose colony-forming cell assays.

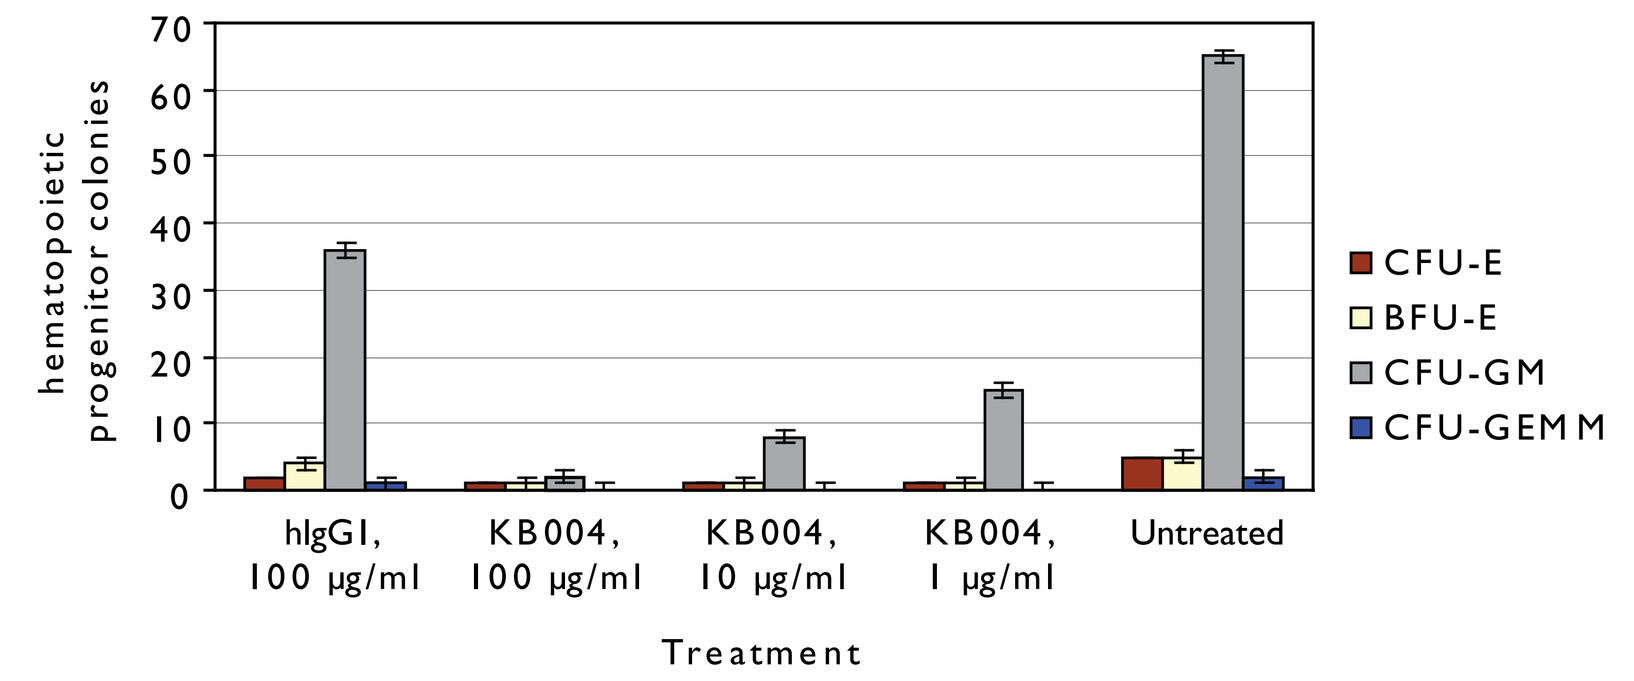
hematopoietic progenitor colonies
hIgG1, 100 µg/ml
KB004, 100 µg/ml
KB004, 10 µg/ml
KB004, 1 µg/ml
Untreated
Treatment
CFU-E
BFU-E
CFU-GM
CFU-GEMM

AML bone marrow cells (2.5x10^5 cells /ml; 95% $EphA3^+$) were plated in MethoCult with or without KB004 or control IgG1 antibody in 35 mm culture dishes. Hematopoietic progenitor-cell colonies (BFU-E, CFU-E, CFU-GM and CFU-GEMM) were counted after 14 days.

Figure 7. KB004 antibody does not affect growth of hematopietic cell colonies from normal human bone marrow CD34+ cells.

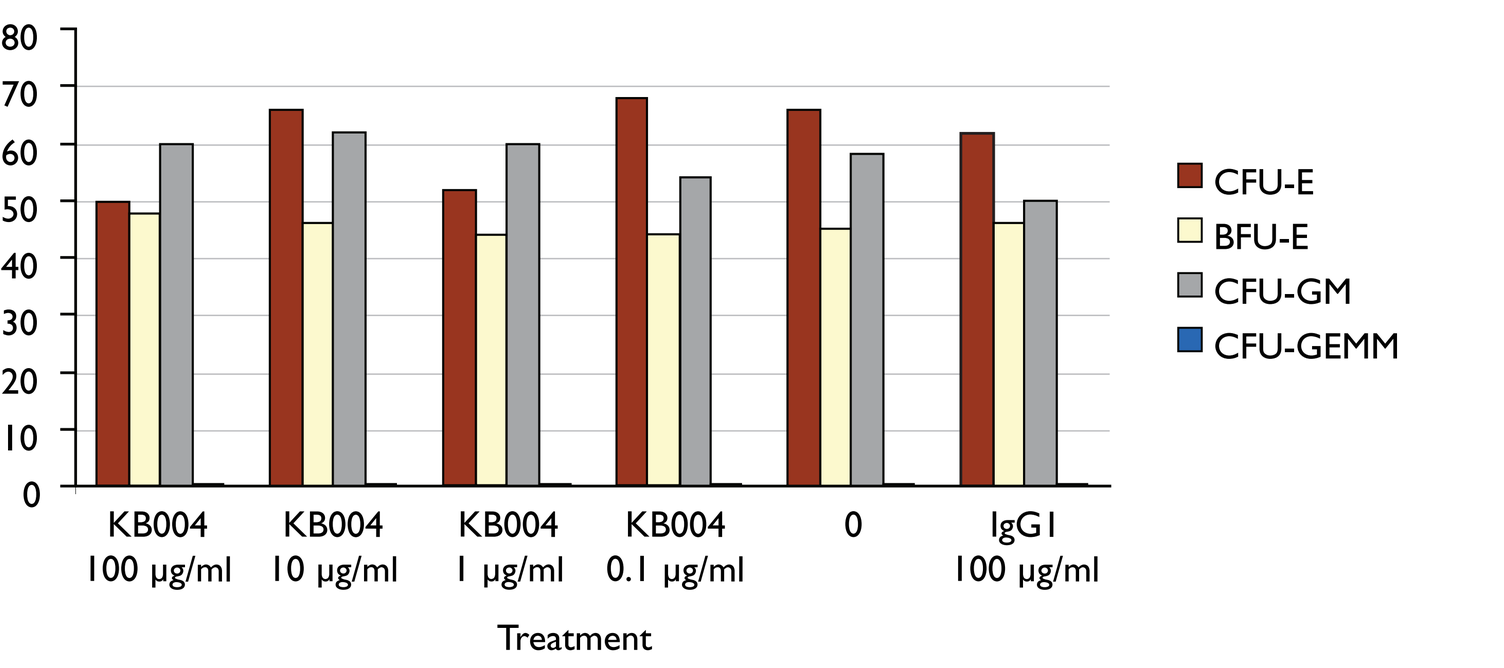
hematopietic progenitor colonies
KB004 100 µg/ml
KB004 10 µg/ml
KB004 1 µg/ml
KB004 0.1 µg/ml
0
IgG1 100 µg/ml
Treatment
CFU-E
BFU-E
CFU-GM
CFU-GEMM

Normal bone marrow CD34+ cells (500 cells/ ml) were plated in MethoCult with or without KB004 or control IgG1 antibody in 35 mm culture dishes. Hematopoietic progenitor cell colonies (BFU-E, CFU-E, CFU-GM and CFU-GEMM) were counted after 14 days.

Table 1. Summary of expression of EphA3 on malignant blast and leukemic stem cells: KB004 kills $EphA3^+$ cells by two independent mechanisms.

	Flow Cytometry Analysis on Bone Marrow Samples					KB004 Activity	
		CD34+ Bone marrow		Leukemic Stem Cells ($CD34^+CD38^-CD123^+$)			
Patient Sample	EphA3+ (% of total cells)	CD34+ (% of total cells)	EphA3+ (% of CD34+ cells)	LSC (% of total cells)	EphA3+ (% of LSC)	% Total Cells Killed by ADCC	% Total Cells Killed by Apoptosis
AML 1	0	0	0	0	N/A	0	0
AML 2	65	0	0	0	N/A	86	86
AML 3	80	80	100	N/D	N/A	47	50
AML 4	95	50	100	25	100	95	79
AML 5	88	9	0	0	N/A	72	79
AML 6	21	23	93	12	99	20	22
AML 7	16	77	21	10	100	20	15
AML 8	24	0	0	0	N/A	22	20
AML 9	38	17	0	0	N/A	40	45
AML 10	41	38	100	0	N/A	50	48
AML 11	55	66	83	0	N/A	65	75
AML 12	14	27	22	0	N/A	20	15
MDS 1	14	21	24	1	100	25	20
MDS 2	14	30	37	3	100	20	19

Primary AML or MDS samples were stained with KB004 antibody, PE-conjugated anti-CD34, PEcy5-conjugated anti-CD38 and APC-conjugated anti-CD123 antibodies for flow cytometry analysis. *In vitro* killing of $EphA3^+$ cells by ADCC and apoptosis correlates with the number of cells expressing detectable levels of EphA3.

CONCLUSIONS

- EphA3 is expressed on the cell surface in approximately 50% of samples from patients with AML, CML, MDS and MPN.
- Humaneered™ anti-EphA3 antibody KB004 has potent cell-killing activity against EphA3-positive hematologic malignancies *in vitro*
 - Direct induction of apoptosis
 - Enhanced ADCC activity
- Cells with leukemic stem cell phenotype ($CD34^+$ $CD38^-$ $CD123^+$) express EphA3.
- KB004 inhibits AML progenitor-cell colony formation and does not inhibit normal human bone marrow progenitor-cell colony formation.
- Opportunity to target leukemic stem cells and blast cells with a potent and selective antibody while sparing normal bone marrow cells.

REFERENCES

Boyd *et al.* (1992) J. Biol. Chem. **267**:3262-3267.

Wicks *et al.* (1992) Proc. Natl. Acad. Sci. U S A. **89**:1611-1615.

Smith *et al.* (1994) J. Biol. Chem. **279**: 9522-9531.

Mori *et al.* (2007) Cytotechnology. **55**:109-14.