Preclinical Studies of RX-5902, a Beta-catenin Modulator, in Triple Negative Breast Cancer

John J. Tentler[1], Julie G Frank[2], DJ Kim[2], Christina George[2], Young B Lee[2], Ely Benaim[2], Aik Choon Tan[1], Jihye Kim[1], Todd M Pitts[1]
Anna Capasso[1] Kyrie L Dailey[1] Jennifer R. Diamond[1]
[1] University of Colorado Comprehensive Cancer Center, Aurora CO [2] Rexahn Inc. Rockville MD

ABSTRACT

Background: RX-5902 (Supinoxin) is a novel anti-cancer compound that targets phosphorylated p68 RNA helicase, a member of the DEAD box family of helicases, affecting upstream and downstream molecules in the Wnt canonical pathway. As a single agent, RX-5902 exhibits strong growth inhibition in both in vitro and in vivo settings. Specifically, RX-5902 enhances survival and tumor growth inhibition in numerous xenograft models, including ovarian, renal and breast. We have previously shown RX-5902 inhibits cell growth in a dose-dependent fashion in the triple-negative breast cancer (TNBC) xenograft MDA-MB231. In the current study, we have expanded our investigation of the therapeutic potential of RX-5902 against TNBC using both in vitro and in vivo preclinical models.

Methods: RX-5902 was provided by Rexahn, Inc. (Rockville, MD). Cell proliferation was measured using the Cell-Titer Glo luminescent cell viability assay (Promega). Apoptosis was assessed using Incucyte Caspase 3/7 Green apoptosis assay (Essenbioscience). Immunoblots of MDA-MB-231 cell line were probed for b-catenin (Cell Signaling). Syngeneic 4T1 murine TNBC mice were obtained from Sippr-BK Laboratory Animal Co (Shanghai, China) and tumor volumes were measured twice a week. When the mean tumor volumes reached ~90 mm^3, mice were randomized and treated with vehicle or RX-5902 PO daily alone or in combination with anti-CTLA4 or anti-PD-1 BIW for 3 weeks. Tumor growth inhibition (TGI) was calculated at Day 25.

Results: A panel of 18 TNBC cell lines were treated with RX-5902 and effects on cell proliferation were assessed. Using 100nM as a cutoff, 14 sensitive lines and 4 resistant lines were identified, with an average IC50 of 56 nM in the sensitive lines. Of these, we chose 2 sensitive lines (MDA-MB-231, HCC1806) and 2 resistant lines (MDA-MB-436 and CAL-120) and assessed induction of apoptosis by the Incucyte caspase activity assay. Robust induction of apoptosis was observed in both sensitive lines (N=3). These lines were then subjected to cell cycle analysis by flow cytometry, which revealed a pronounced G2/M cell cycle arrest and aneuploidy following exposure to RX-5902. Western blot analysis of the MDA-MB-231 cell line showed decreases in the Wnt pathway-related protein nuclear β-catenin in doses ranging from 20 nM to 200 nM. Finally, the therapeutic efficacy of RX-5902 was assessed as a single agent and in combination with two immune-oncology agents in the treatment of the TNBC 4T1 animal model. RX-5902 as a single agent showed dose dependency in the 4T1 model, and when given in combination with either anti-CTLA4 or anti-PD1 showed an additive effect ($p<0.001$). All the treatments were well-tolerated and no severe body weight loss was observed in this study.

Conclusions: RX-5902 showed efficacy against several in vitro and in vivo preclinical models of TNBC. RX-5902 resulted in G2/M arrest and induced apoptosis in sensitive TNBC cell lines and decreases in nuclear beta-catenin. In vivo, RX-5902 demonstrated additive anti-tumor effects when combined with either anti-CTLA4 or anti-PD1 immunotherapies. Together, these finding indicate that RX-5902 may have important clinical implications for the treatment of TNBC. A phase 2a clinical study in metastatic TNBC is ongoing.

MATERIALS AND METHODS

Drugs: RX-5902 was provided by Rexahn, Inc. (Rockville, MD). Anti-CTLA4 and anti-PD-1 checkpoint inhibitors were obtained from XXX and the University of Colorado Hospital Pharmacy, respectively.
Cell proliferation was measured using the Cell-Titer Glo luminescent cell viability assay (Promega). **Apoptosis** was assessed using Incucyte Caspase 3/7 Green apoptosis assay (Essenbioscience). **Immunoblots** of MDA-MB-231 cell line were probed for beta-catenin (Cell Signaling).
Animal Models: Syngeneic 4T1 murine TNBC mice were obtained from Sippr-BK Laboratory Animal Co (Shanghai, China) and tumor volumes were measured twice a week. When the mean tumor volumes reached ~90 mm^3, mice were randomized and treated with vehicle or RX-5902 PO daily alone or in combination with anti-CTLA4 or anti-PD-1 BIW for 3 weeks. Tumor growth inhibition (TGI) was calculated at Day 25.
Humanized Sirpα mice developed from the BALB/cRag2-/-IL2Rγc-/- (BRG) strain were used to enhance engraftment. Newborn pups were humanized through transplantation of approximately 1x105 CD34+ cells purified from umbilical cord blood. Mice were evaluated for chimerism at 8 and 12 weeks. At 16 weeks, tumor tissue from established PDX models were implanted on the right and left flanks of humanized mice.
When average tumor size reached a volume of ~150-300 mm3, mice were randomized into vehicle or nivolumab treatment groups according to %chimerism. Tumor size was evaluated twice weekly by caliper measurements: tumor volume= (length × width2) × 0.52.

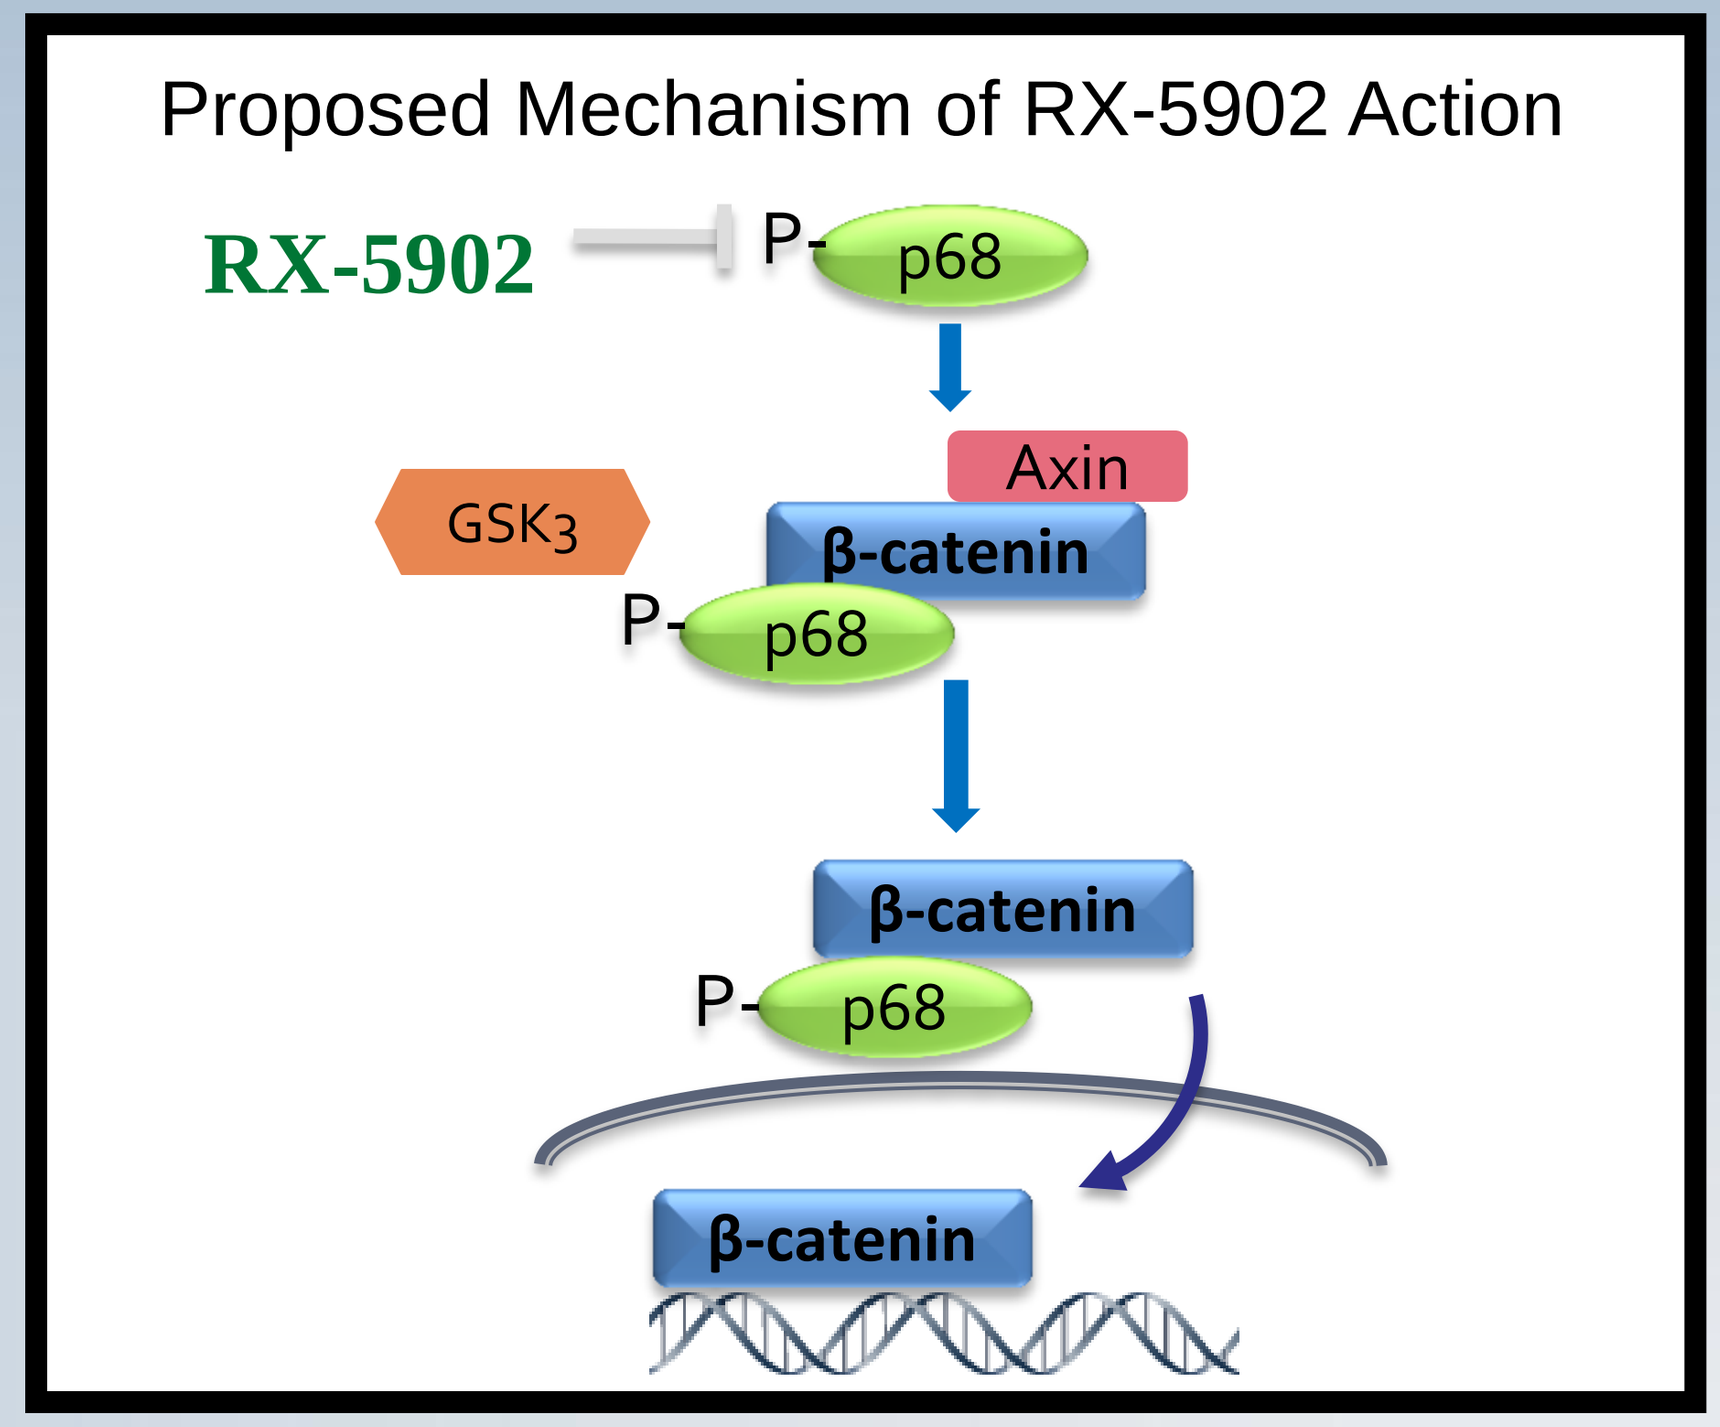
Proposed Mechanism of RX-5902 Action
RX-5902
P-
p68
Axin
GSK3
β-catenin
β-catenin
β-catenin

Figure 1. Mechanism of RX-5902 action

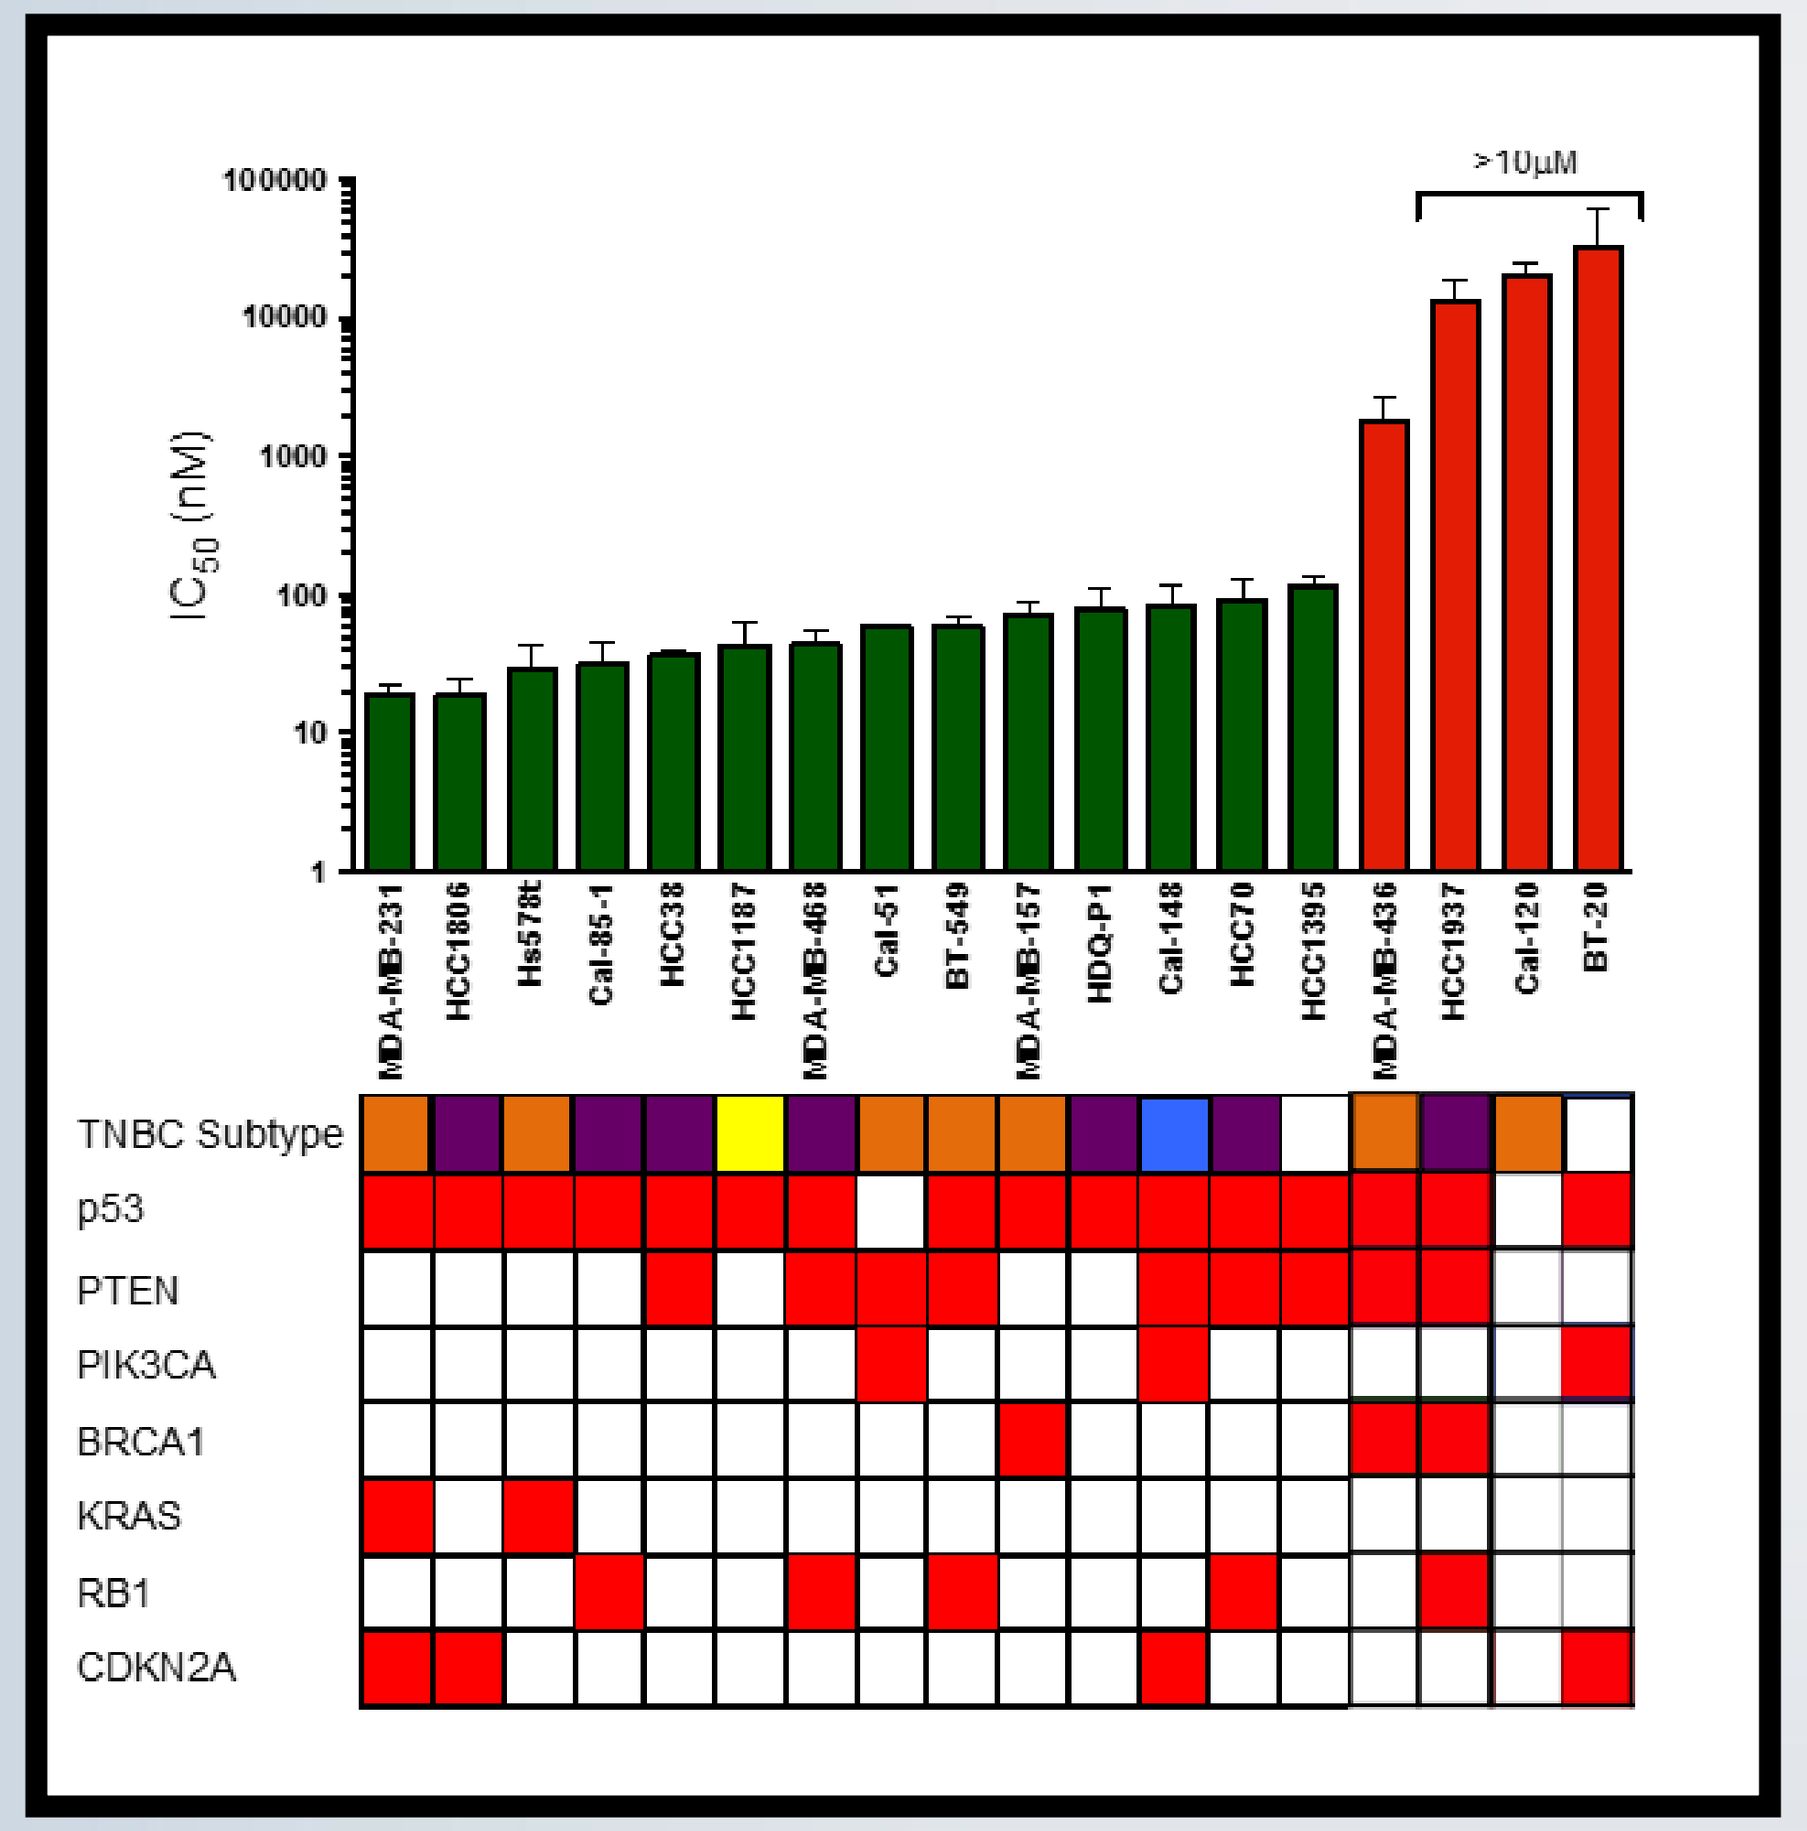
IC_{50} (nM)
>10µM
MDA-MB-231
HCC1806
Hs578t
Cal-85-1
HCC38
HCC1187
MDA-MB-468
Cal-51
BT-549
MDA-MB-157
HDQ-P1
Cal-148
HCC70
HCC1395
MDA-MB-436
HCC1937
Cal-120
BT-20
TNBC Subtype
p53
PTEN
PIK3CA
BRCA1
KRAS
RB1
CDKN2A

Figure 2. IC50 values for a panel of TNBC cell lines exposed to RX-5902 for 72 hrs. as measured by Cell Titer Glo assay

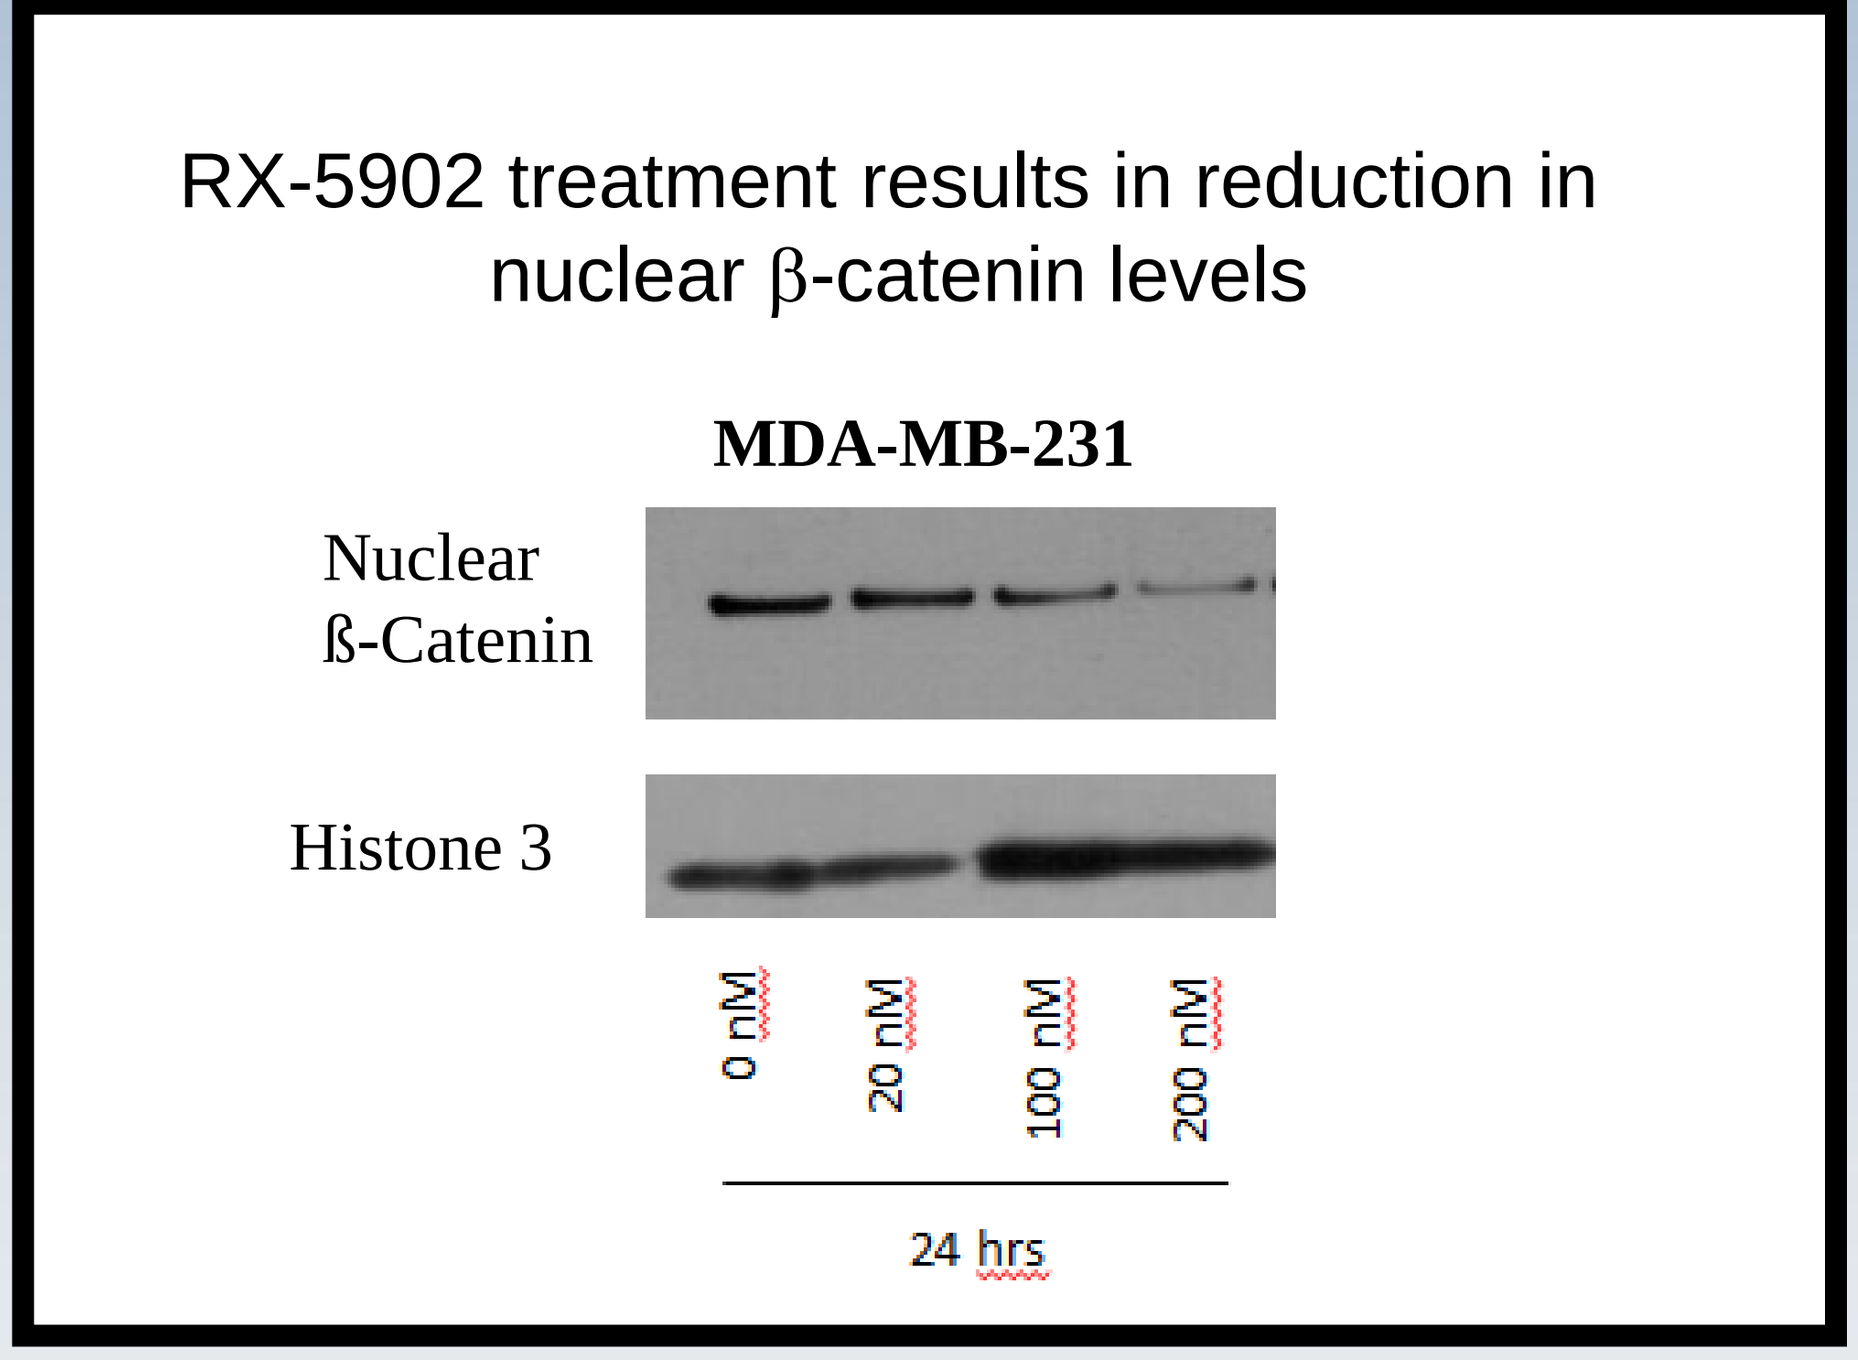
RX-5902 treatment results in reduction in nuclear β-catenin levels
MDA-MB-231
Nuclear ß-Catenin
Histone 3
0 nM
20 nM
100 nM
200 nM
24 hrs

Figure 3. Immunoblot analysis of MDA231 cell line nuclear extracts from cell exposed to increasing doses of RX-5902

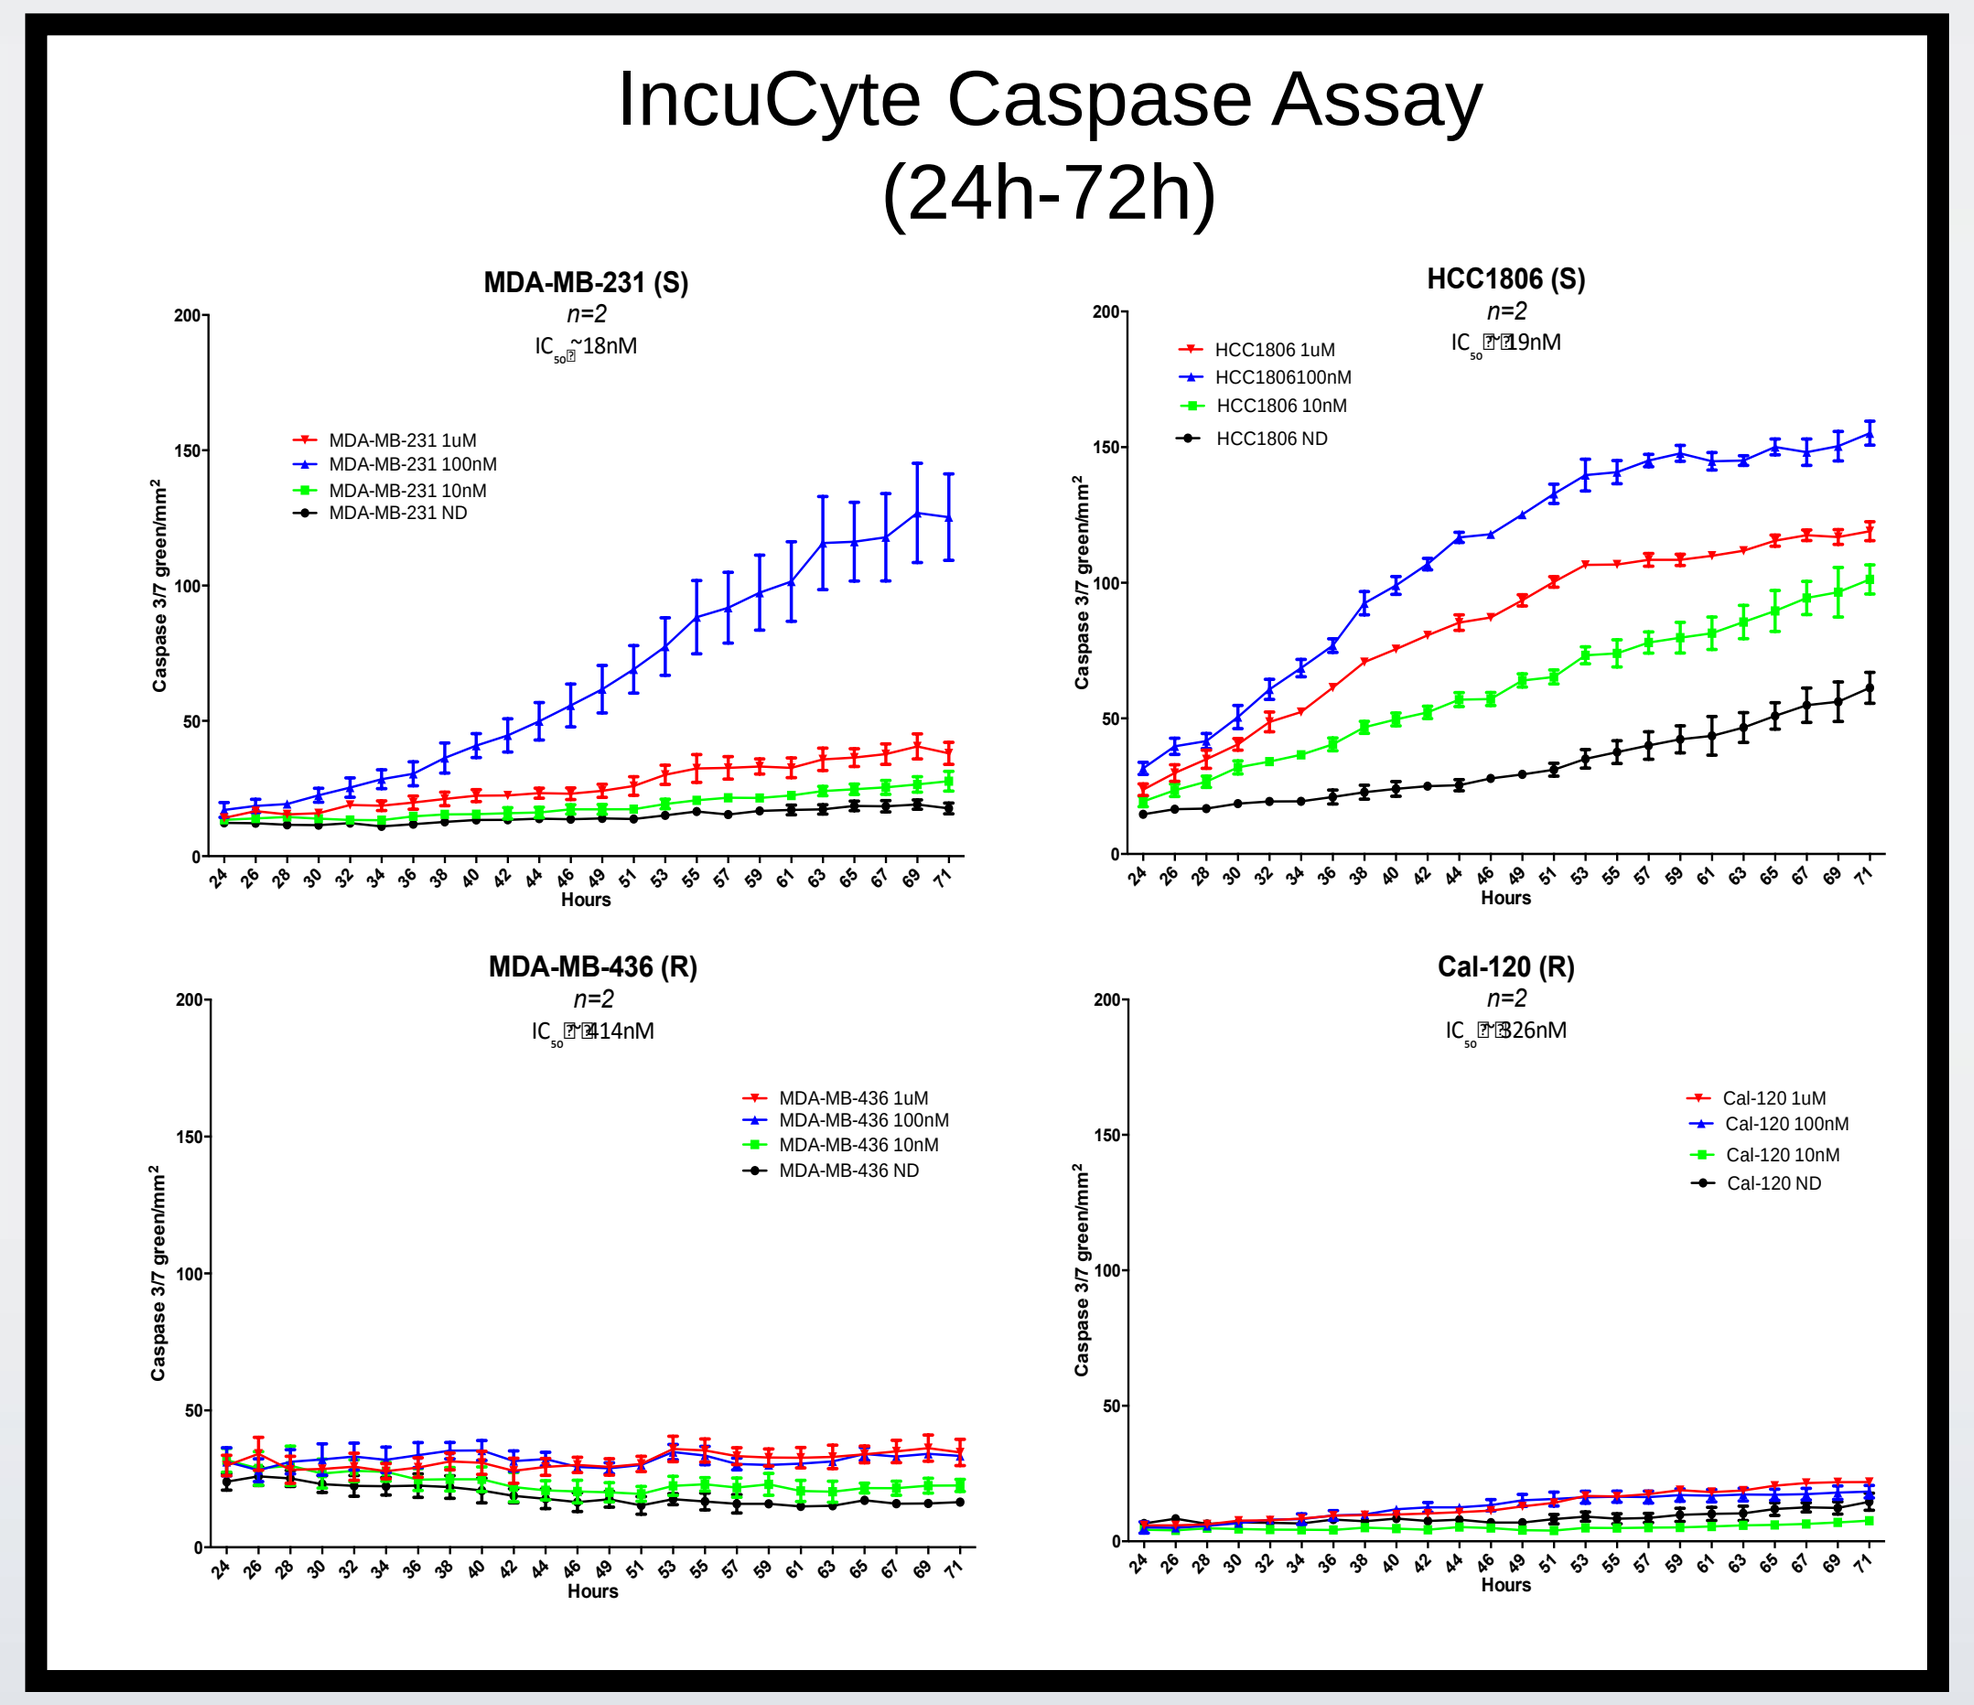
IncuCyte Caspase Assay
(24h-72h)
MDA-MB-231 (S)
HCC1806 (S)
MDA-MB-436 (R)
Cal-120 (R)
Hours

Figure 4. Caspase 3/7 activity over time as measured by IncuCyte for the indicated doses of RX-5902

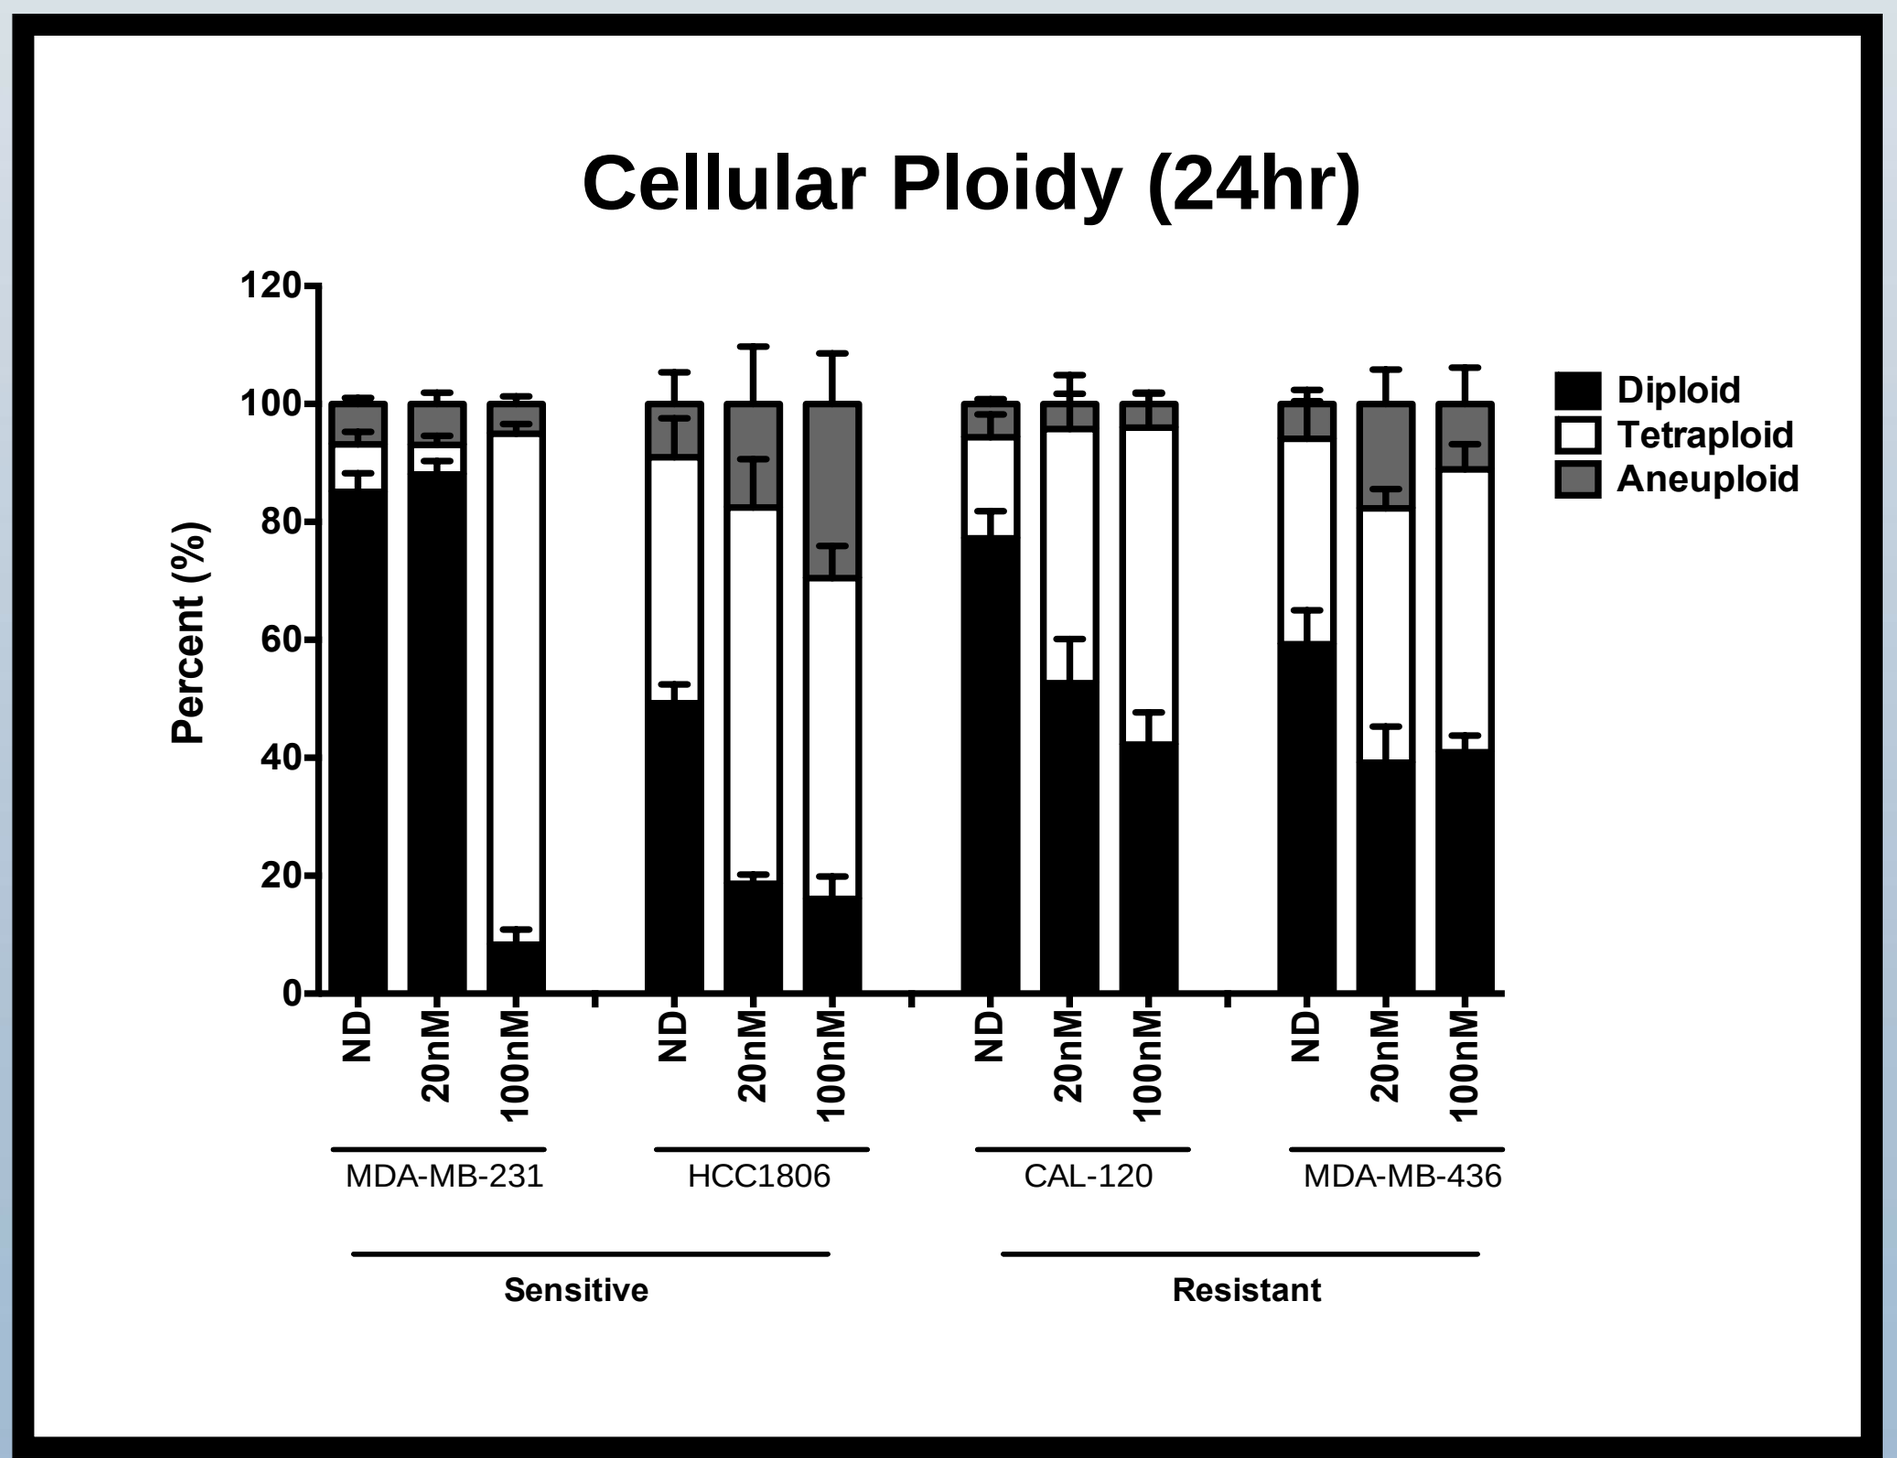
Cellular Ploidy (24hr)
Percent (%)
Diploid
Tetraploid
Aneuploid
ND
20nM
100nM
MDA-MB-231
HCC1806
CAL-120
MDA-MB-436
Sensitive
Resistant

Figure 5. Measurement of ploidy by IncuCyte at the Indicated doses of RX-5902 after 24 hrs treatment

Additive Effects of RX-5902 and Anti-CTLA4 in Tumor Growth Inhibition in 4T1 Triple Negative Breast Cancer Model

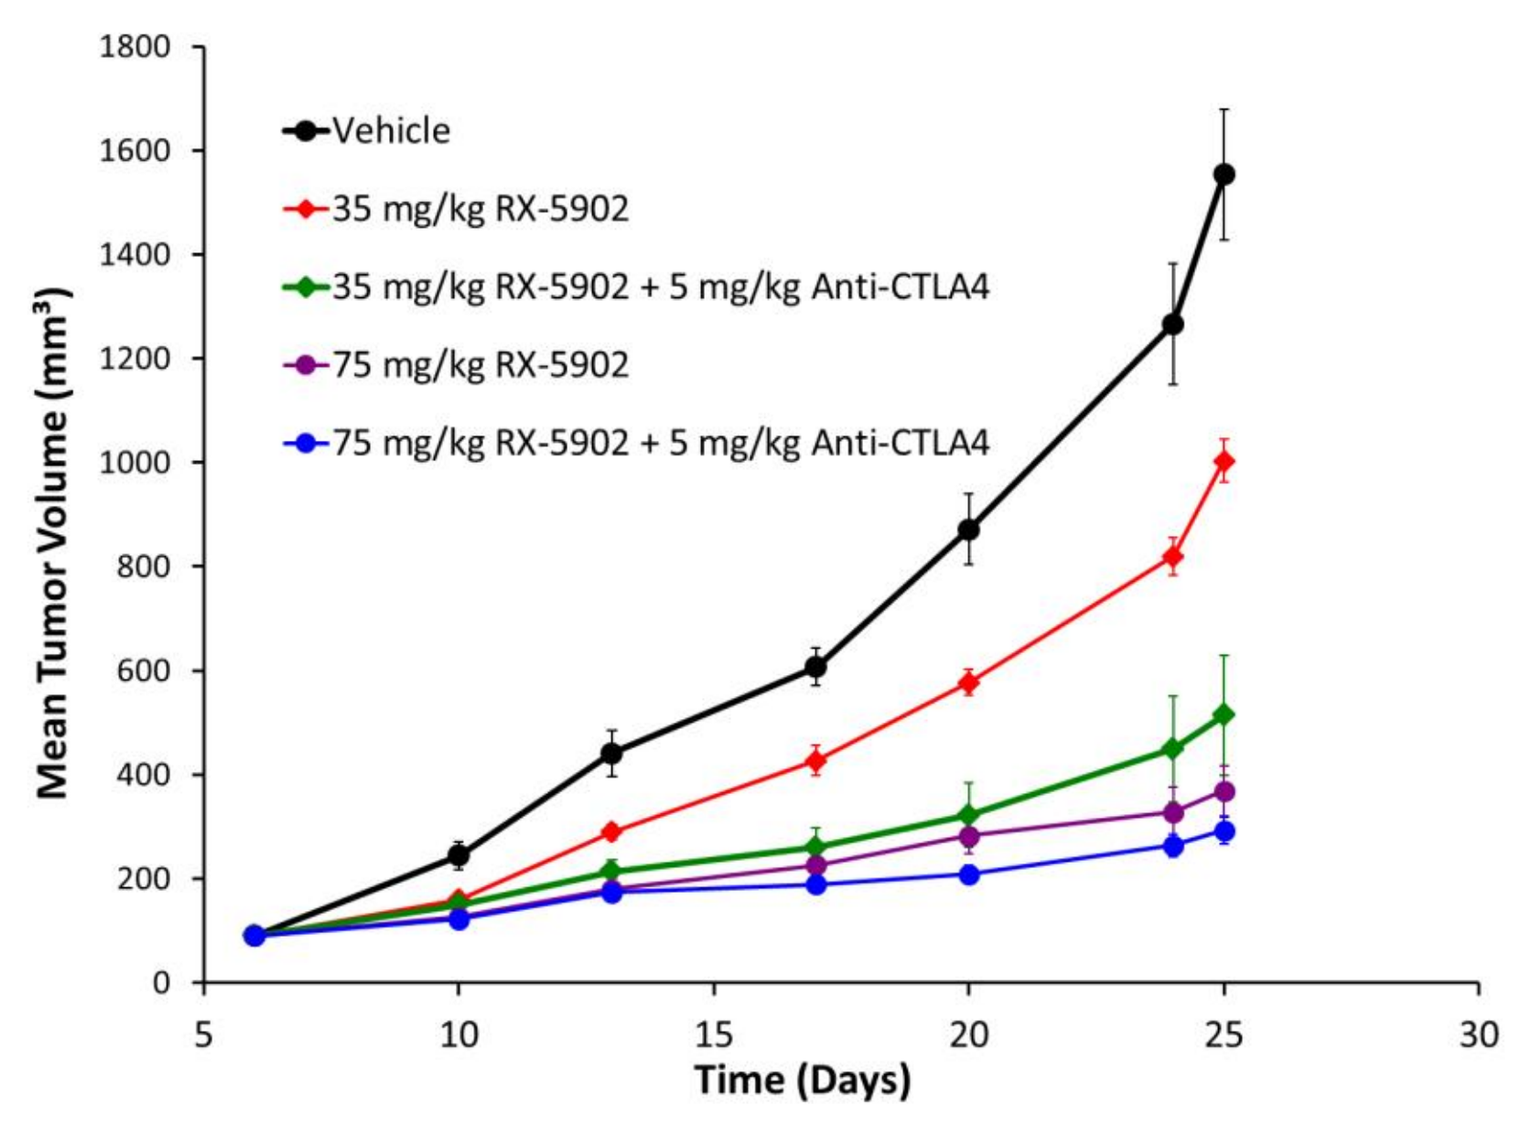
Vehicle
35 mg/kg RX-5902
35 mg/kg RX-5902 + 5 mg/kg Anti-CTLA4
75 mg/kg RX-5902
75 mg/kg RX-5902 + 5 mg/kg Anti-CTLA4
Mean Tumor Volume (mm^3)
Time (Days)

Treatment Group	TGI Day 21
Vehicle	-
35 mg/kg RX-5902	35%
35 mg/kg RX-5902 + anti-CTLA4	67%
75 mg/kg RX-5902	76%
75 mg/kg RX-5902 + anti-CTLA4	81%

Figure 6. Significant dose dependent inhibition of tumor growth was observed with RX-5902 and additive effect in combination with anti-CTLA4 immunotherapy

Additive Effects of RX-5902 and Anti-PD-1 in Tumor Growth Inhibition in 4T1Triple Negative Breast Cancer Model

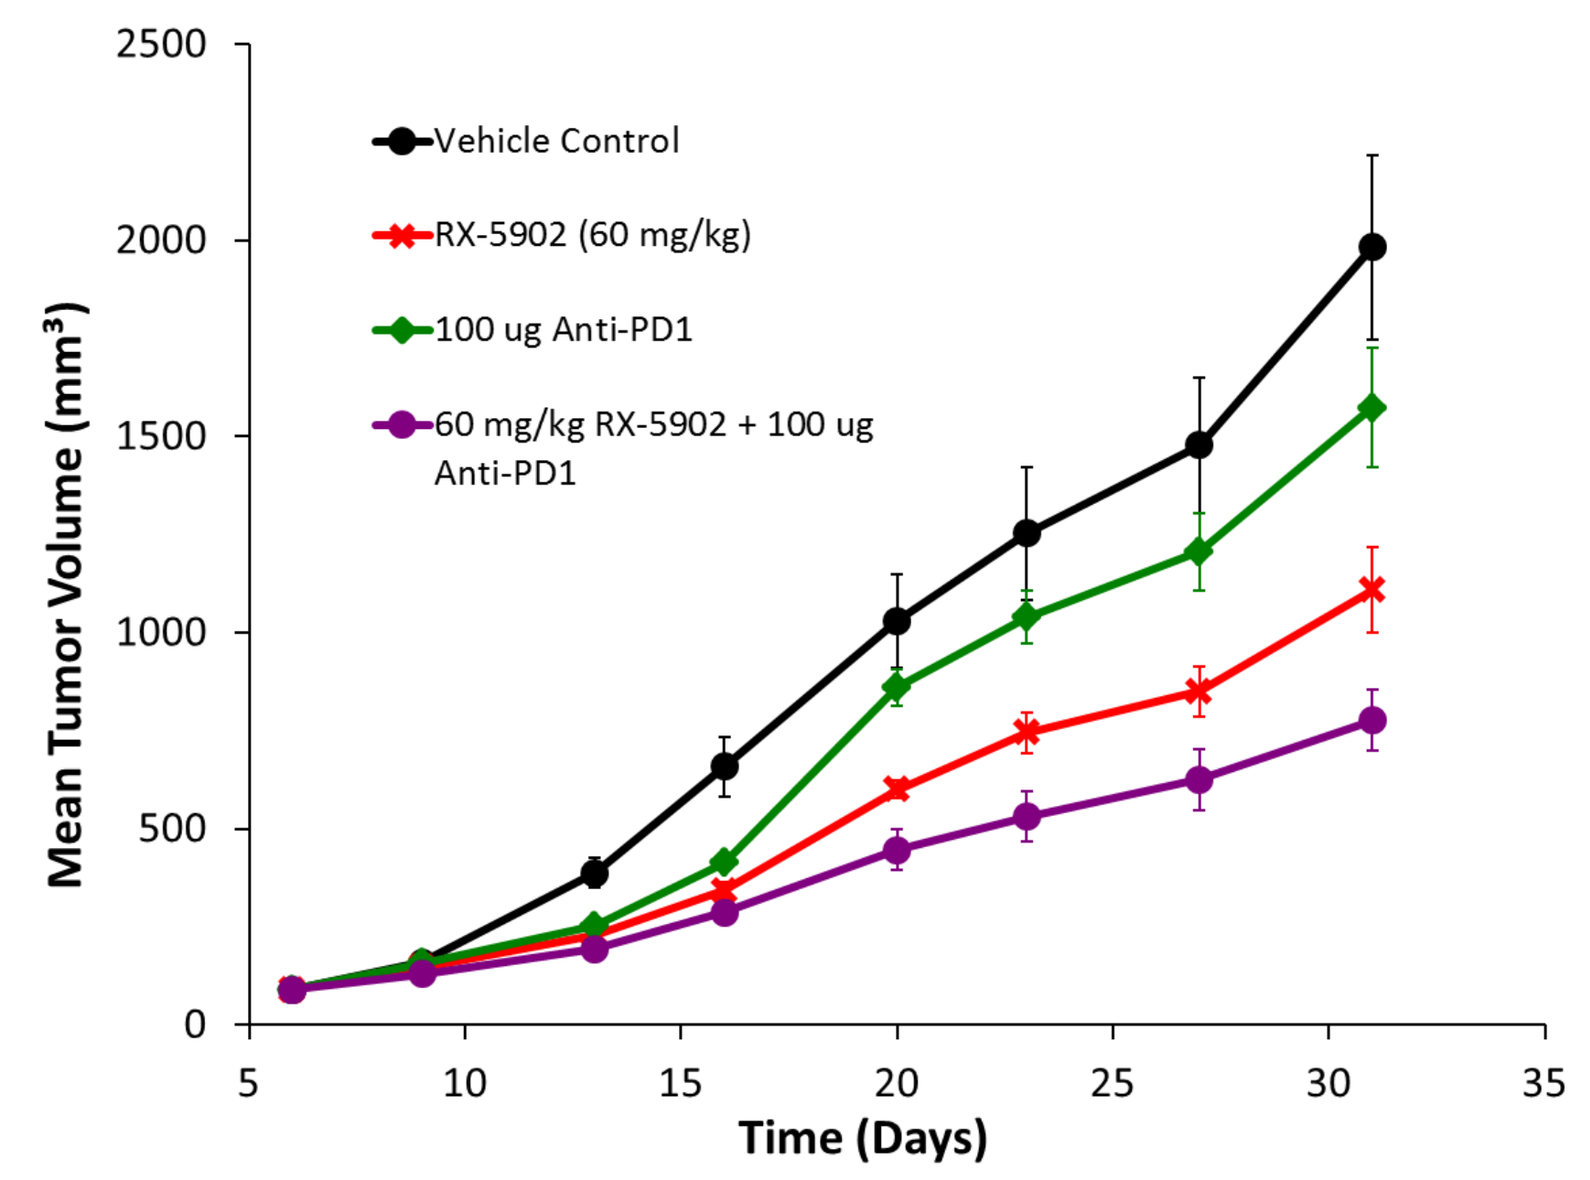
Vehicle Control
RX-5902 (60 mg/kg)
100 ug Anti-PD1
60 mg/kg RX-5902 + 100 ug Anti-PD1
Mean Tumor Volume (mm^3)
Time (Days)

Treatment Group	TGI Day 25
Vehicle	-
RX-5902	46%
anti-PD-1 RMP1-14	21%
RX-5902 + anti-PD-1	64%

Figure 7. Dose dependent inhibition of tumor growth was observed with RX-5902 and additive effect in combination with anti-PD-1 immunotherapy

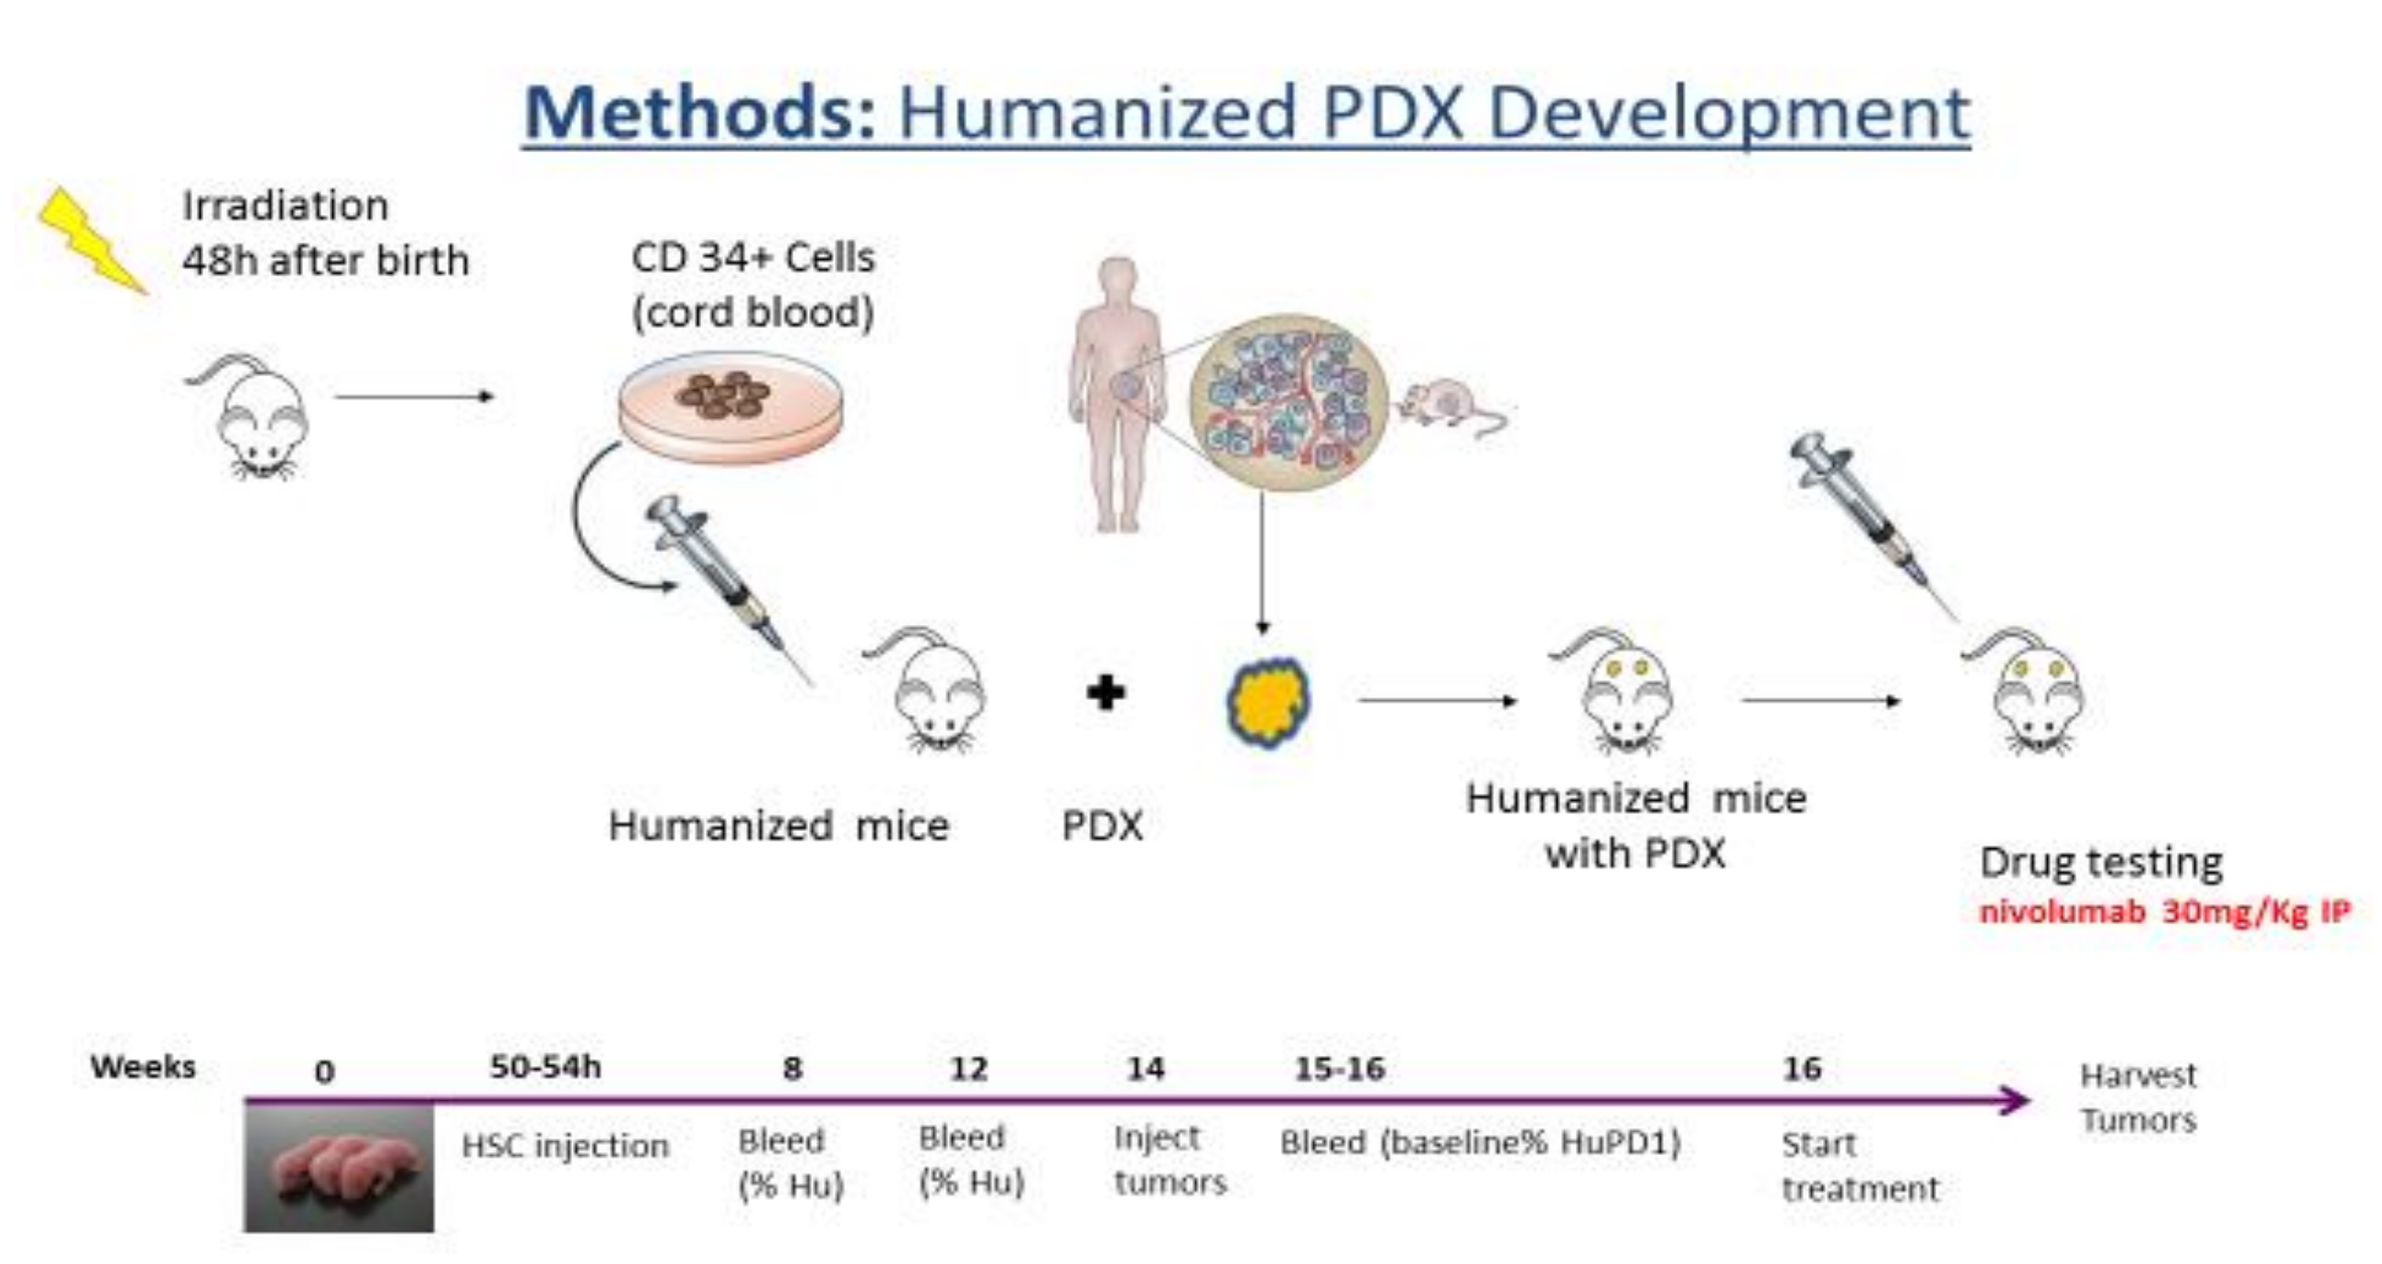
Methods: Humanized PDX Development
Irradiation 48h after birth
CD 34+ Cells (cord blood)
Humanized mice
PDX
Humanized mice with PDX
Drug testing
nivolumab 30mg/Kg IP
Weeks
0
50-54h
8
12
14
15-16
16
HSC injection
Bleed (% Hu)
Bleed (% Hu)
Inject tumors
Bleed (baseline% HuPD1)
Start treatment
Harvest Tumors

Figure 8. Schema for generation of humanized mice for testing combinations with immunotherapies with cell lines or PDX samples

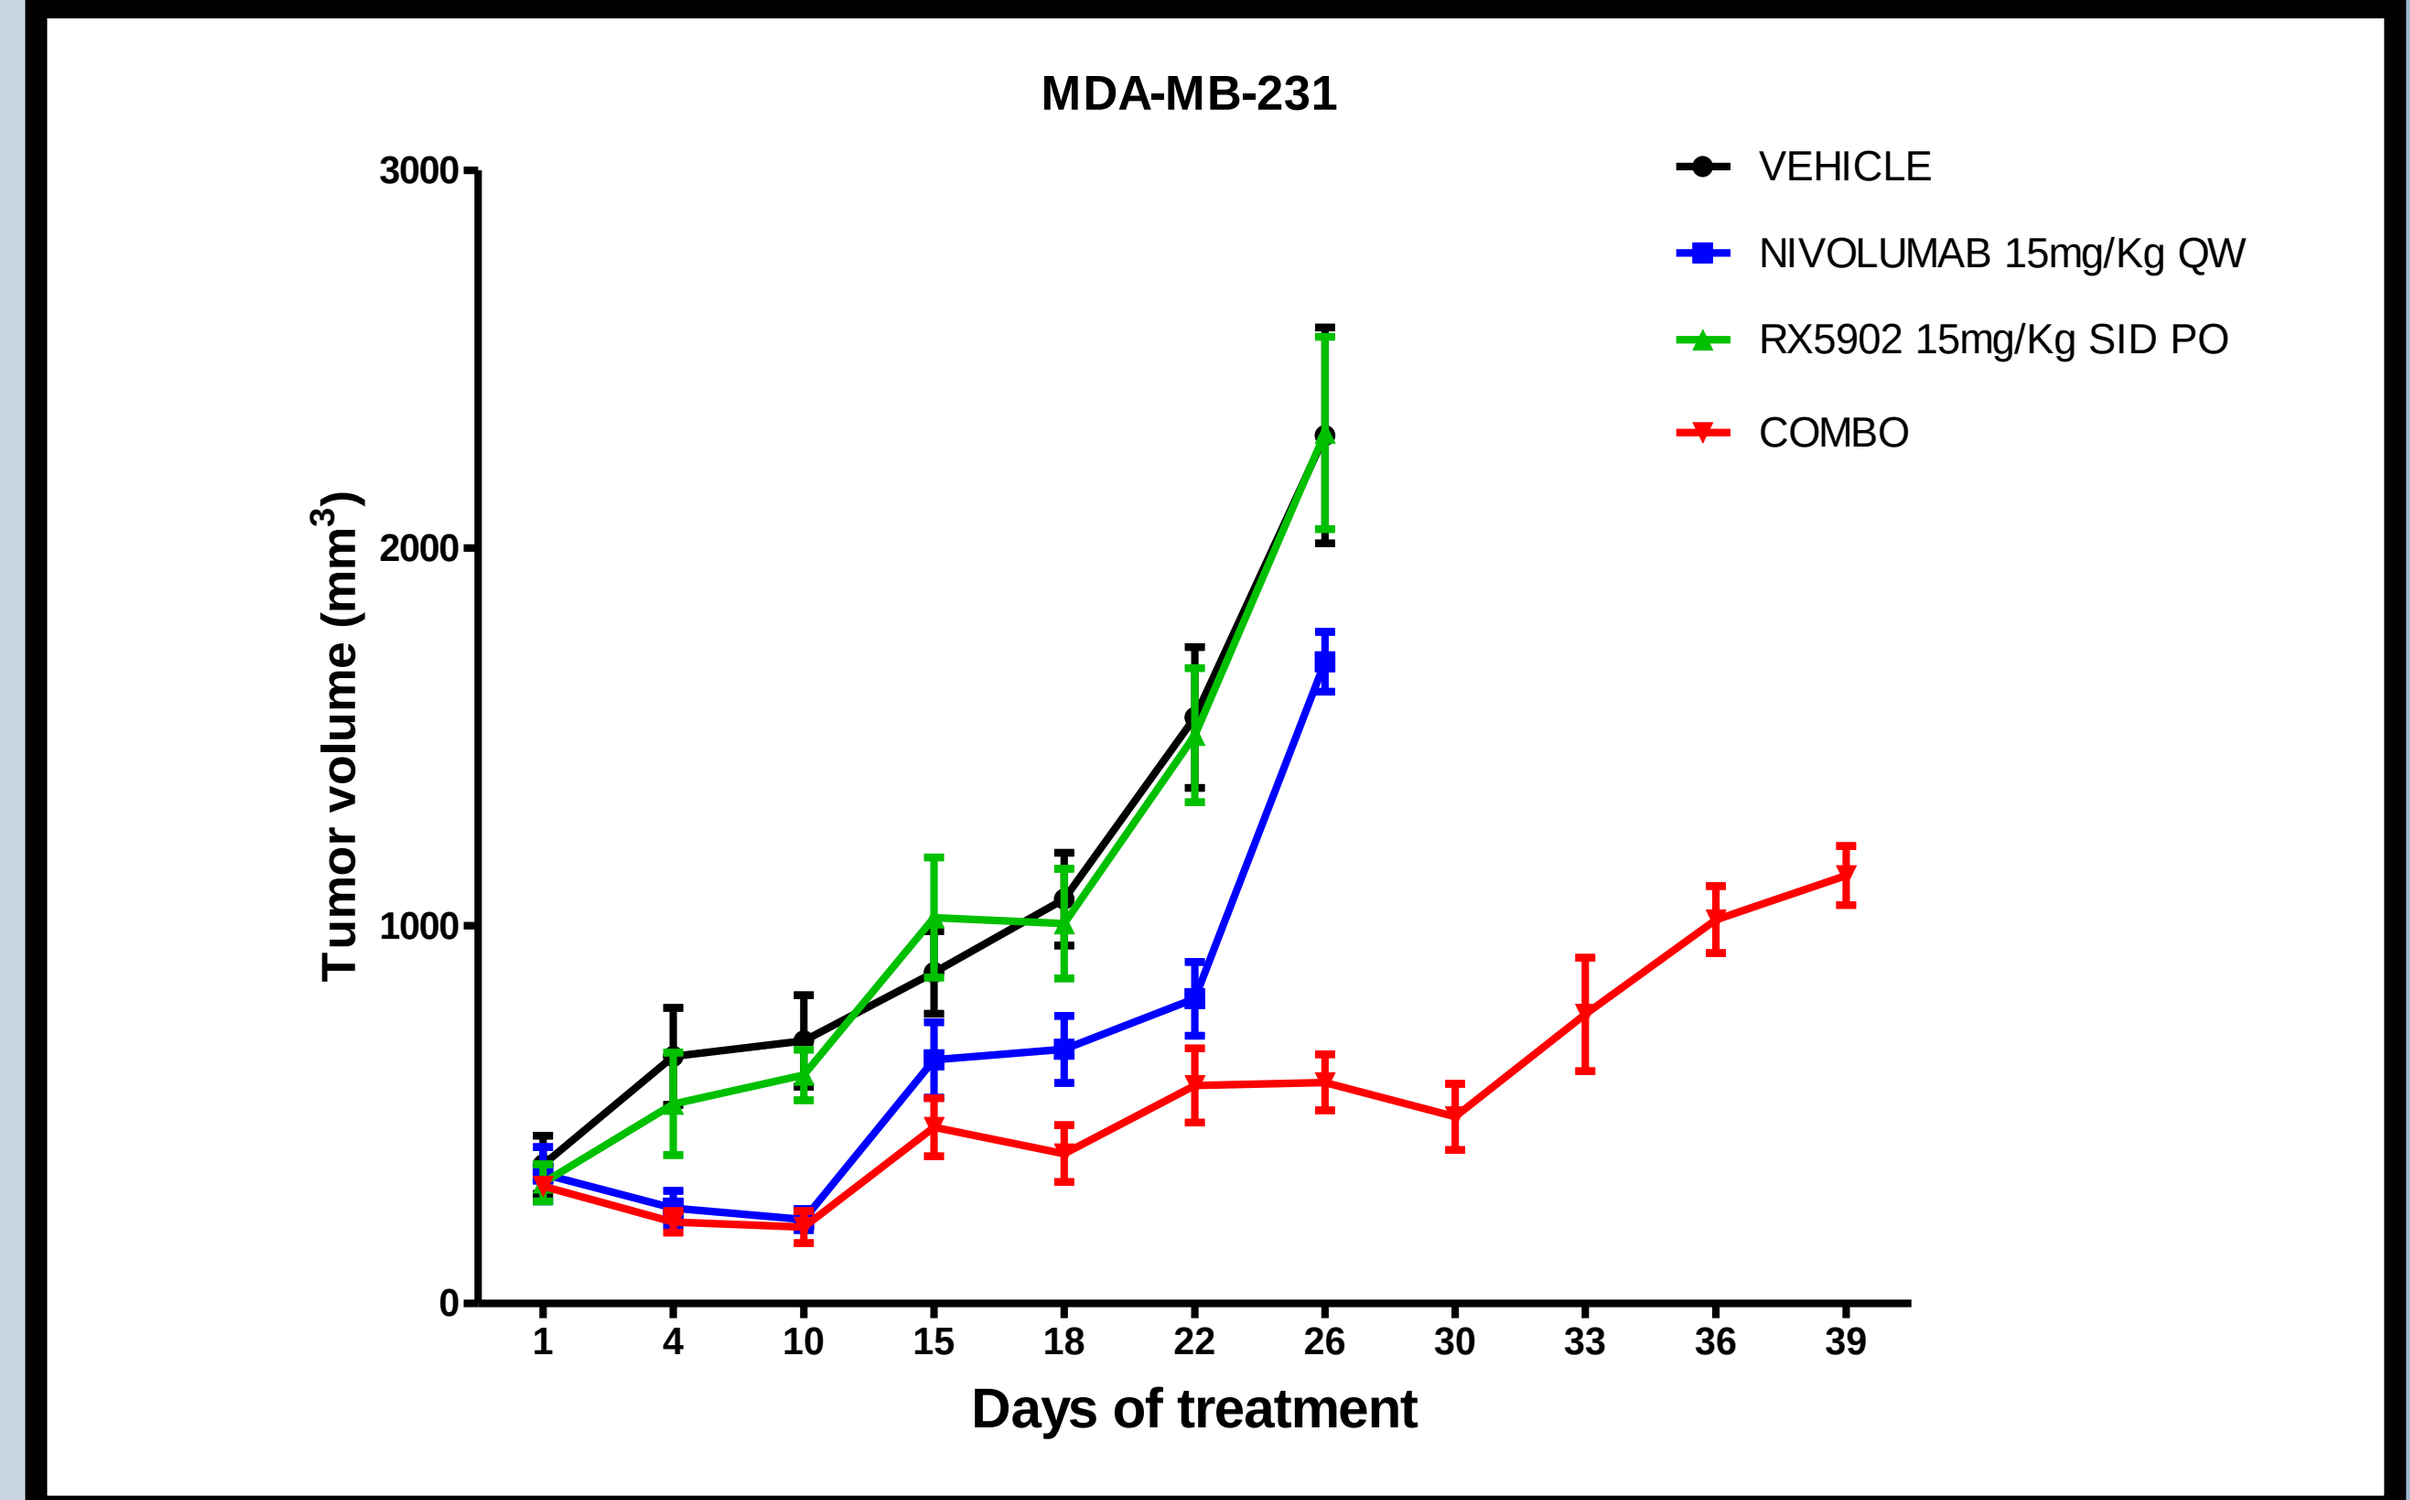
MDA-MB-231
VEHICLE
NIVOLUMAB 15mg/Kg QW
RX5902 15mg/Kg SID PO
COMBO
Tumor volume (mm^3)
Days of treatment

Figure 9. Tumor growth curves of humanized mice bearing MDA-MB-231 TNBC cell line xenograft tumors at the indicated doses

CONCLUSIONS

- RX-5902 demonstrated efficacy against several in vitro and in vivo preclinical models of TNBC
- RX-5902 resulted in G2/M arrest and induced apoptosis in sensitive TNBC cell lines and decreases in nuclear beta-catenin
- In vivo, RX-5902 demonstrated additive anti-tumor effects when combined with either anti-CTLA4 or anti-PD1 immunotherapies in 4T1 syngeneic mouse models
- In a humanized mouse model bearing MDA-MB-231 cell line xenograft tumors, we observed significant tumor growth inhibition when RX-5902 is combined with the anti-PD-1 checkpoint inhibitor nivolumab
- Together, these finding indicate that RX-5902 may have important clinical implications for the treatment of TNBC

FUNDING

Funding for this project was provided through an MTA with Rexahn Inc