A Clinical Practice Assessment on *Clostridium difficile* Infection

JOVANA LUBARDA, PHD; JOHN MAEGLIN, Medscape LLC, New York, United States **DEB MATHEWS, PHARMD; TRUDI DELK, PHARMD;** Synthetic Biologics, Maryland, United States
EDWARD J. SEPTIMUS, MD, Texas A&M College of Medicine, Internal Medicine, Houston, United States

BACKGROUND

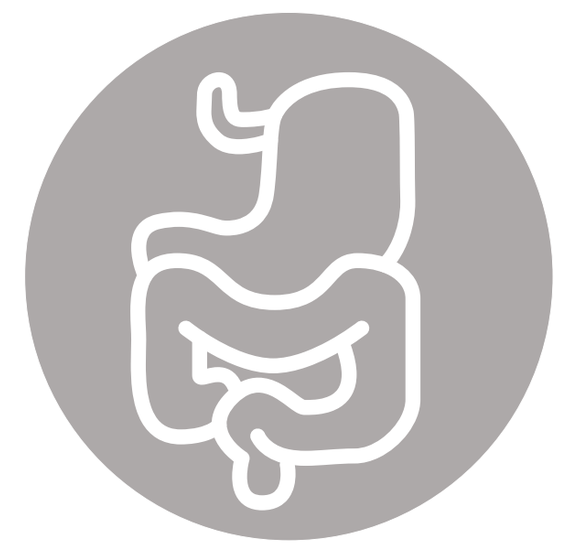

The goal of this study was to assess physicians' current practice patterns in prevention and management of *Clostridium difficile* infection (CDI).

METHODS

- A 25-question clinical practice assessment survey was made available to infectious disease (ID) specialists without monetary compensation or charge
- Questions evaluated knowledge, competence, and barriers related to CDI, such as current and emerging strategies for limiting risk and achieving optimal outcomes related to antimicrobial use
- The survey launched on a website dedicated to continuous professional development on October 27, 2017. Data were collected until January 16, 2018
- Respondent confidentiality was maintained, and responses were de-identified and aggregated prior to analyses

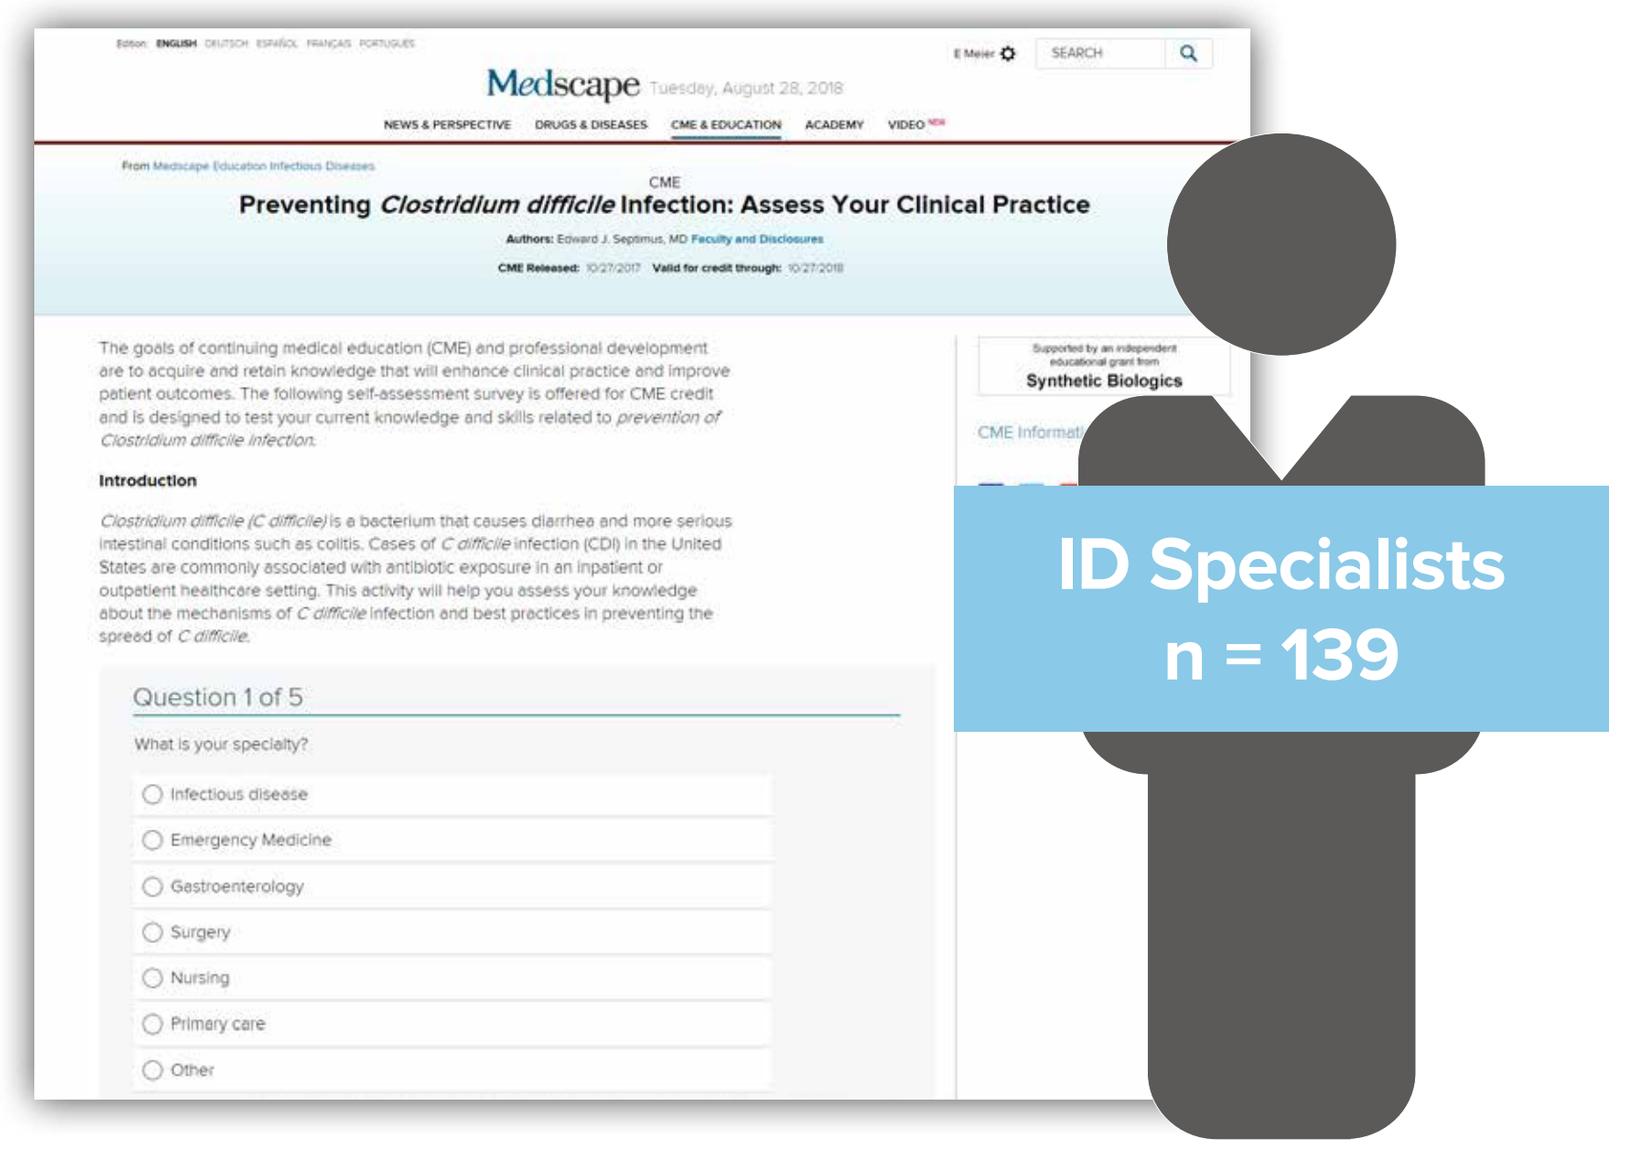
Medscape
Preventing *Clostridium difficile* Infection: Assess Your Clinical Practice
ID Specialists
n = 139

RESULTS

INCIDENCE OF CDI

A total of 76% were not aware of the incidence of CDI in the United States

Which of the following is an accurate statement about CDI in the United States?

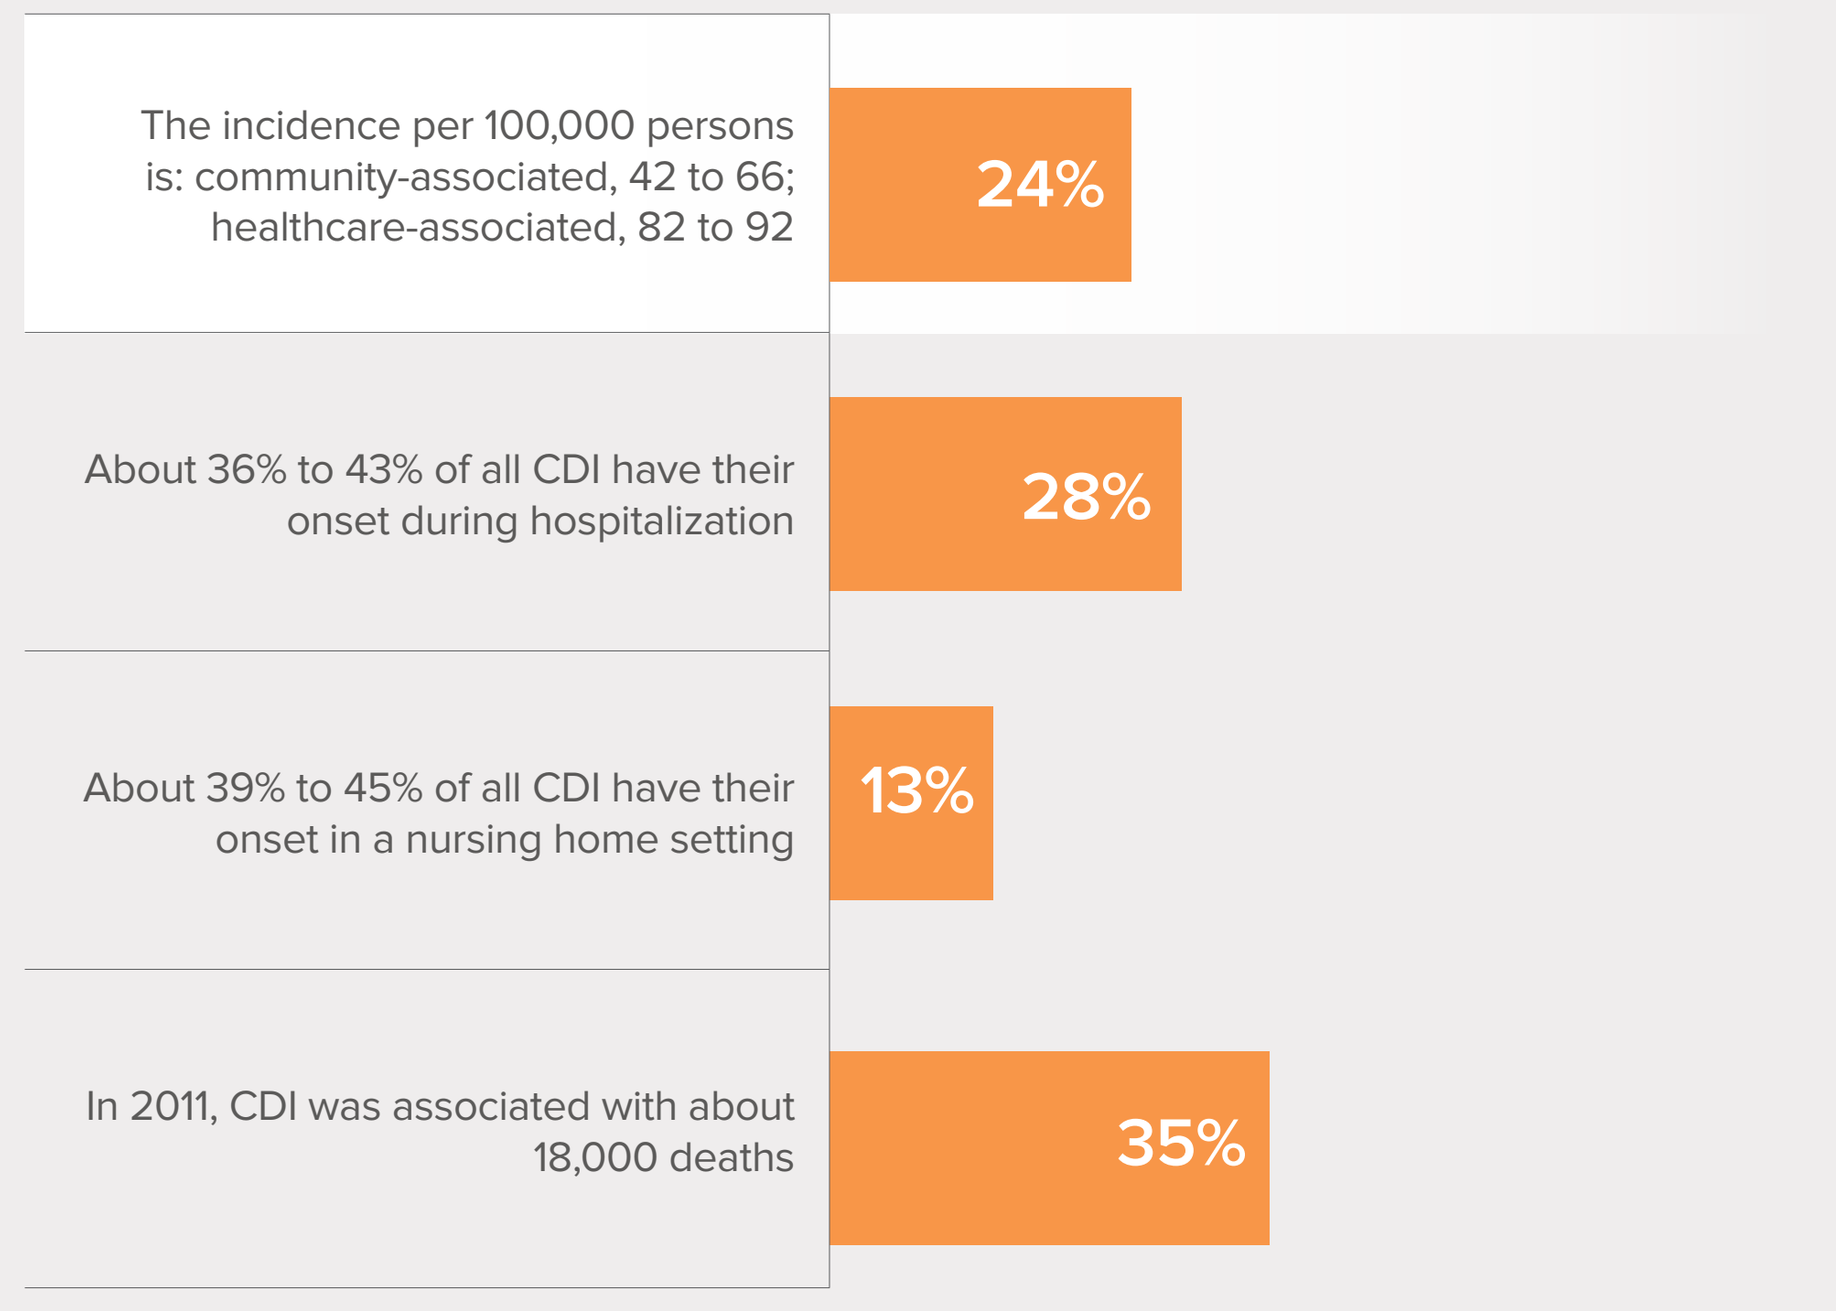
The incidence per 100,000 persons is: community-associated, 42 to 66; healthcare-associated, 82 to 92
24%
About 36% to 43% of all CDI have their onset during hospitalization
28%
About 39% to 45% of all CDI have their onset in a nursing home setting
13%
In 2011, CDI was associated with about 18,000 deaths
35%

About 34% had more than 20 cases of CDI in their practice in the past year

Approximately how many cases of CDI have occurred in your practice in the past year?

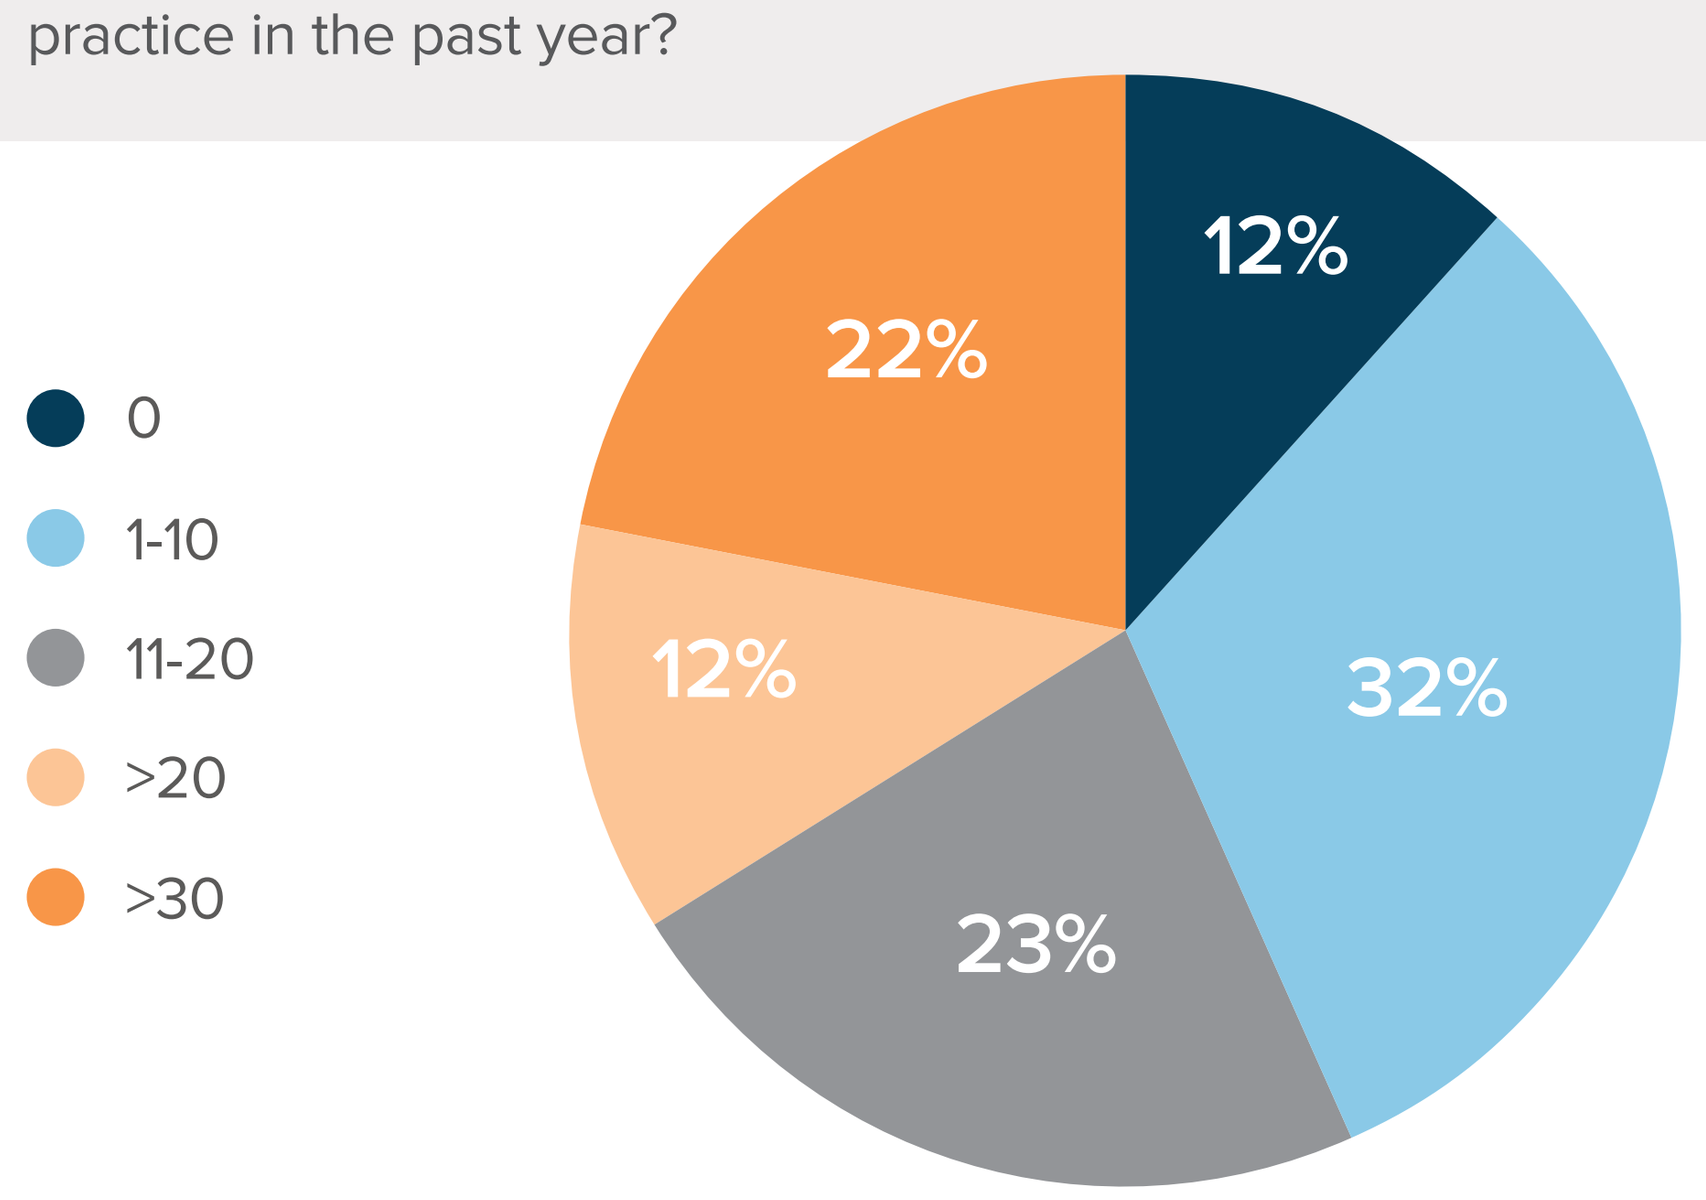
0
1-10
11-20
>20
>30
12%
32%
23%
12%
22%

RISK FACTORS FOR CDI

Nearly all (96%) physicians correctly identified the type or class of antibiotic most closely associated with development of CDI

The use of which of the following antibiotic or class of antibiotics is most closely associated with the development of CDI?

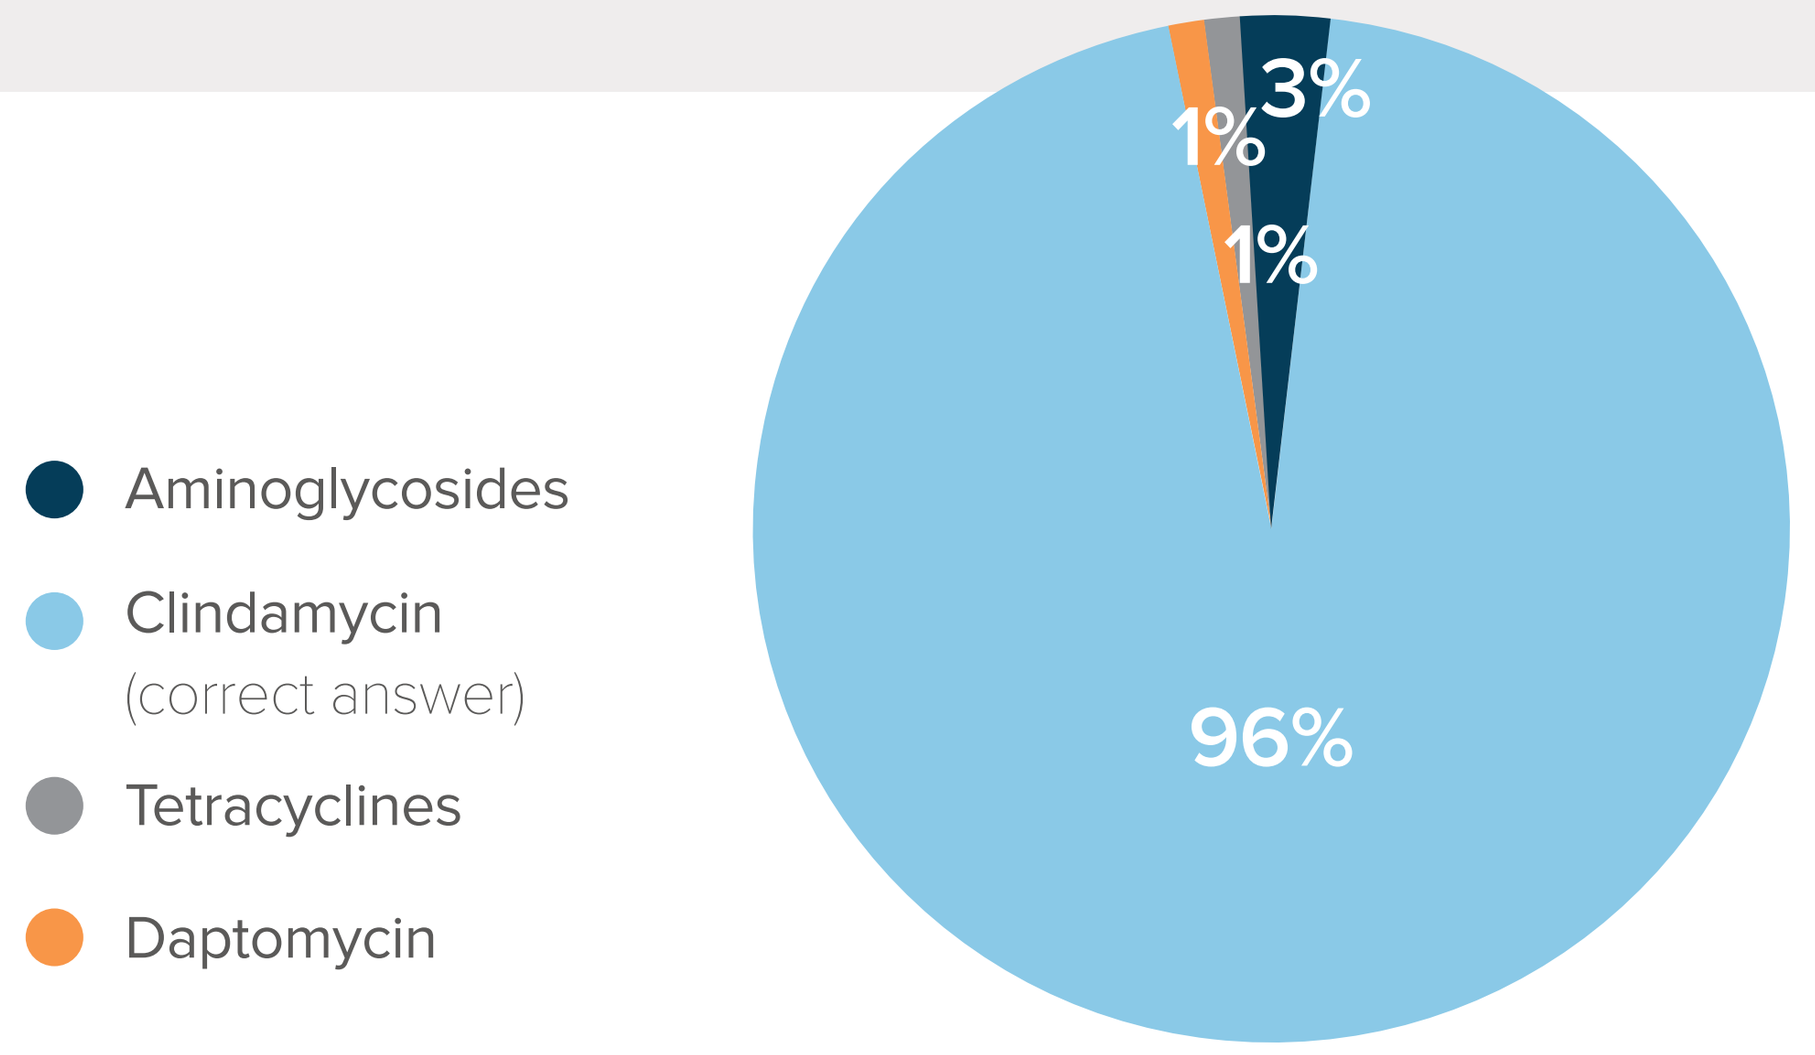
Aminoglycosides
Clindamycin
(correct answer)
Tetracyclines
Daptomycin
3%
1%
1%
96%

Only 22% reported they were very confident in recognizing host risk factors for CDI

Two events are required for the development of CDI: Disruption of the fecal microbiota (typically via use of antibiotics) and ingestion of spores via the fecal-oral route. Host factors also have a role in a person's risk for CDI. How confident are you in recognizing host risk factors for CDI?

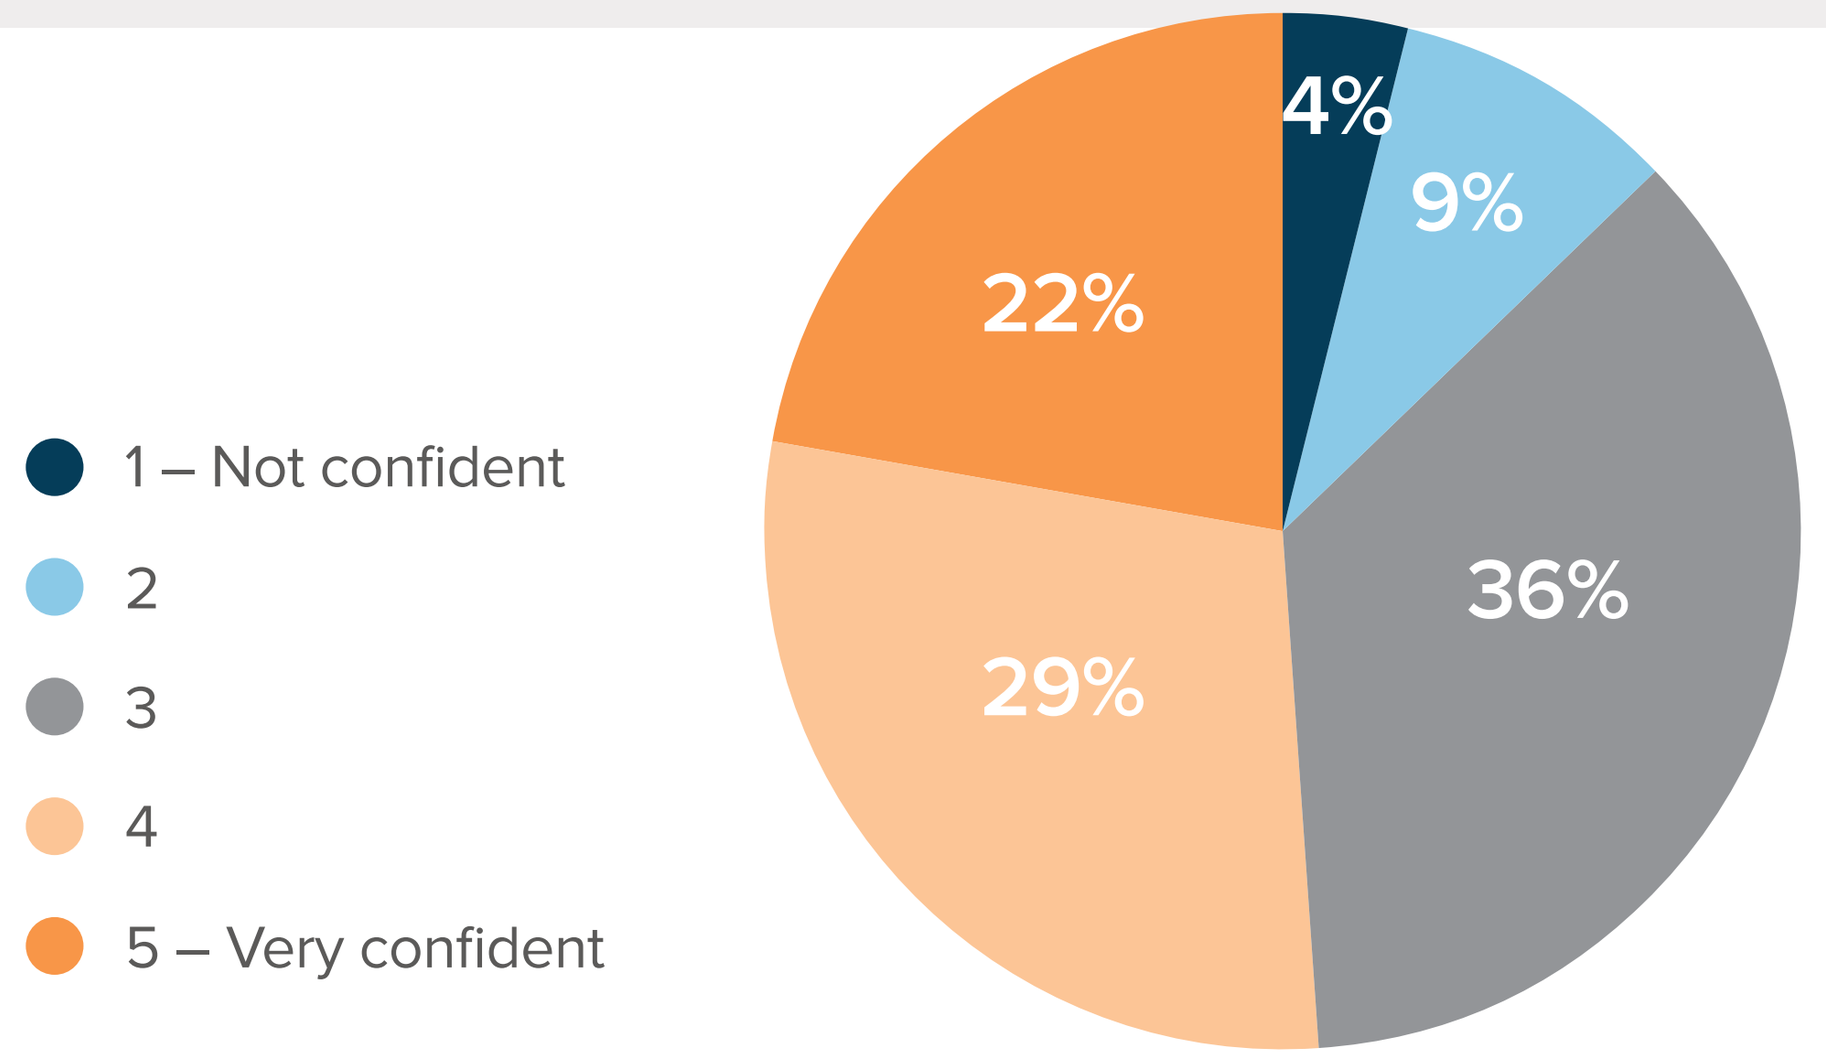
1 – Not confident
2
3
4
5 – Very confident
4%
9%
36%
29%
22%

39% were not aware of the relationship between the gut microbiome and CDI

Which of the following is not an accurate statement about the relationship between gut microbiota and CDI?

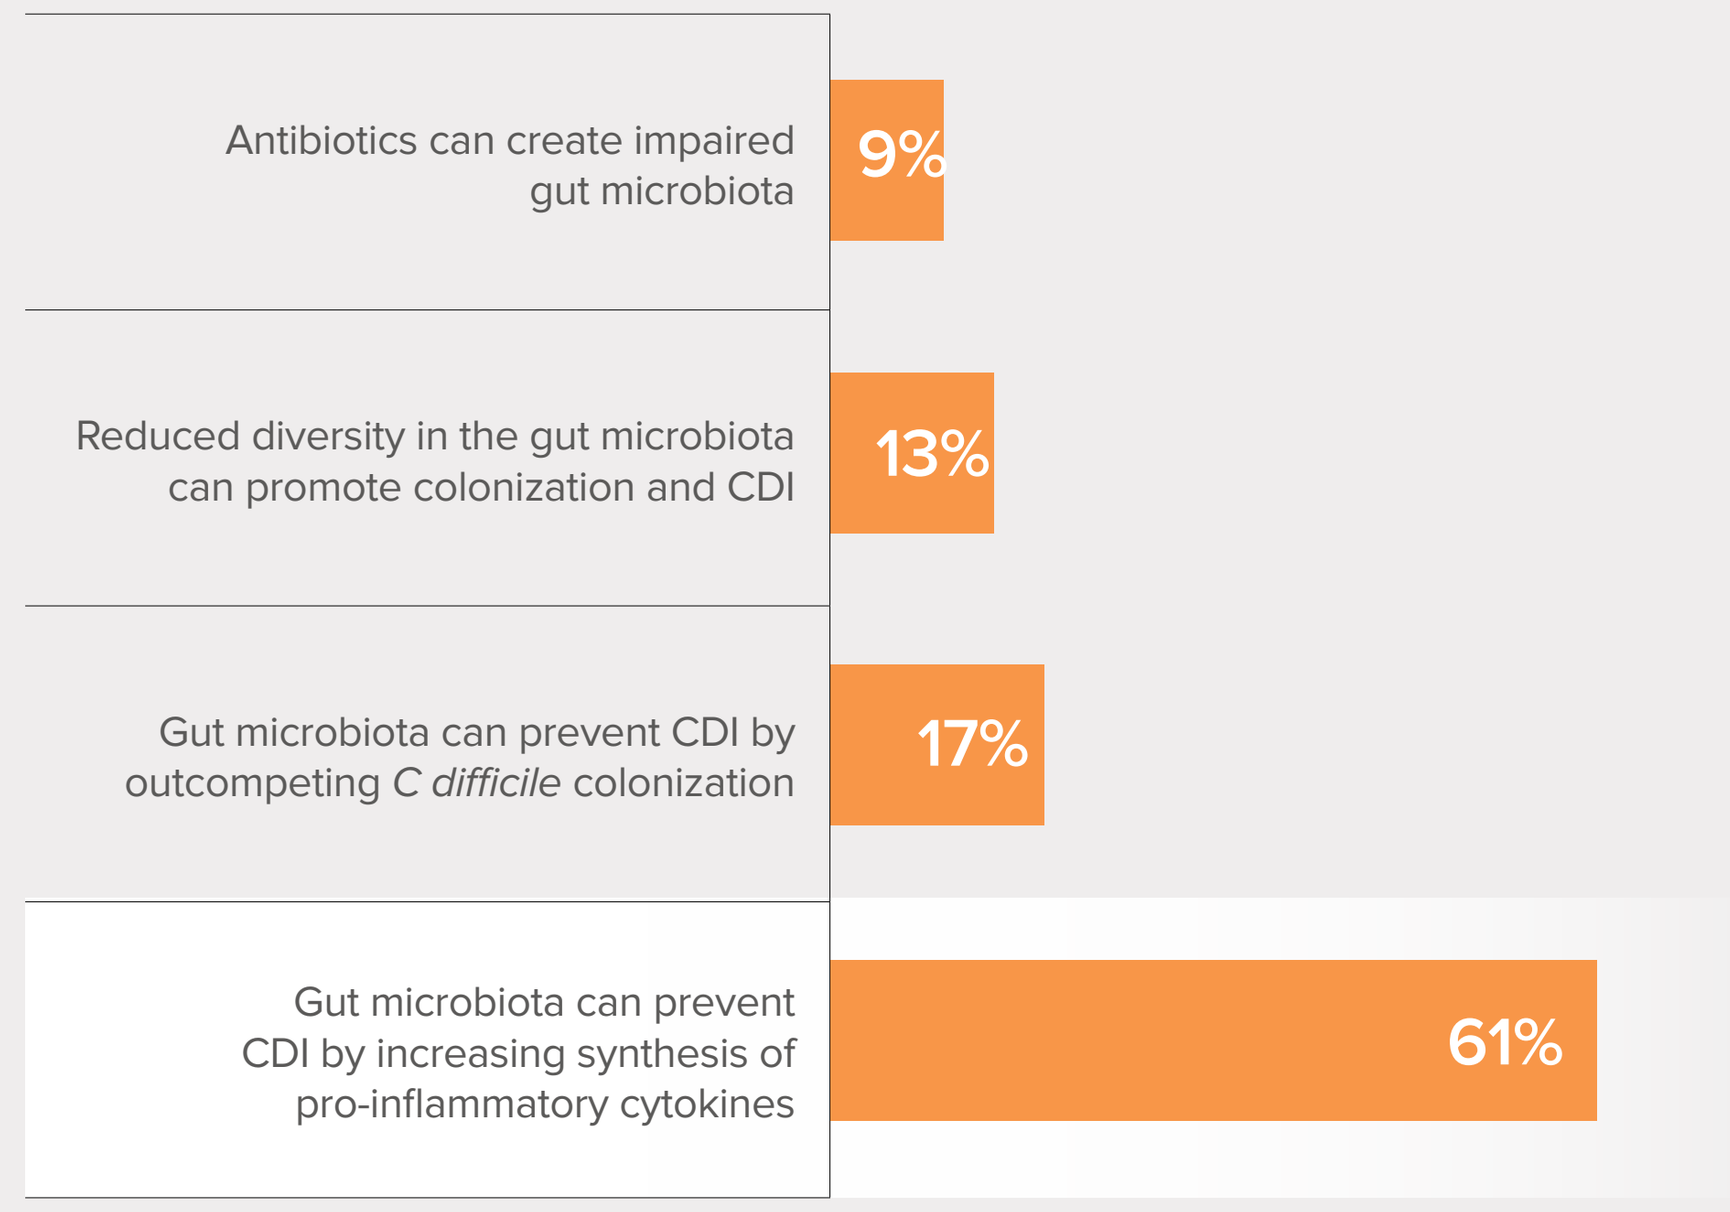
Antibiotics can create impaired gut microbiota
9%
Reduced diversity in the gut microbiota can promote colonization and CDI
13%
Gut microbiota can prevent CDI by outcompeting *C difficile* colonization
17%
Gut microbiota can prevent CDI by increasing synthesis of pro-inflammatory cytokines
61%

DIAGNOSIS OF CDI

38% use a polymerase chain reaction-based method for CDI diagnosis; 36% use a 2-step method combining different test types

What does your institution use for the diagnosis of CDI?

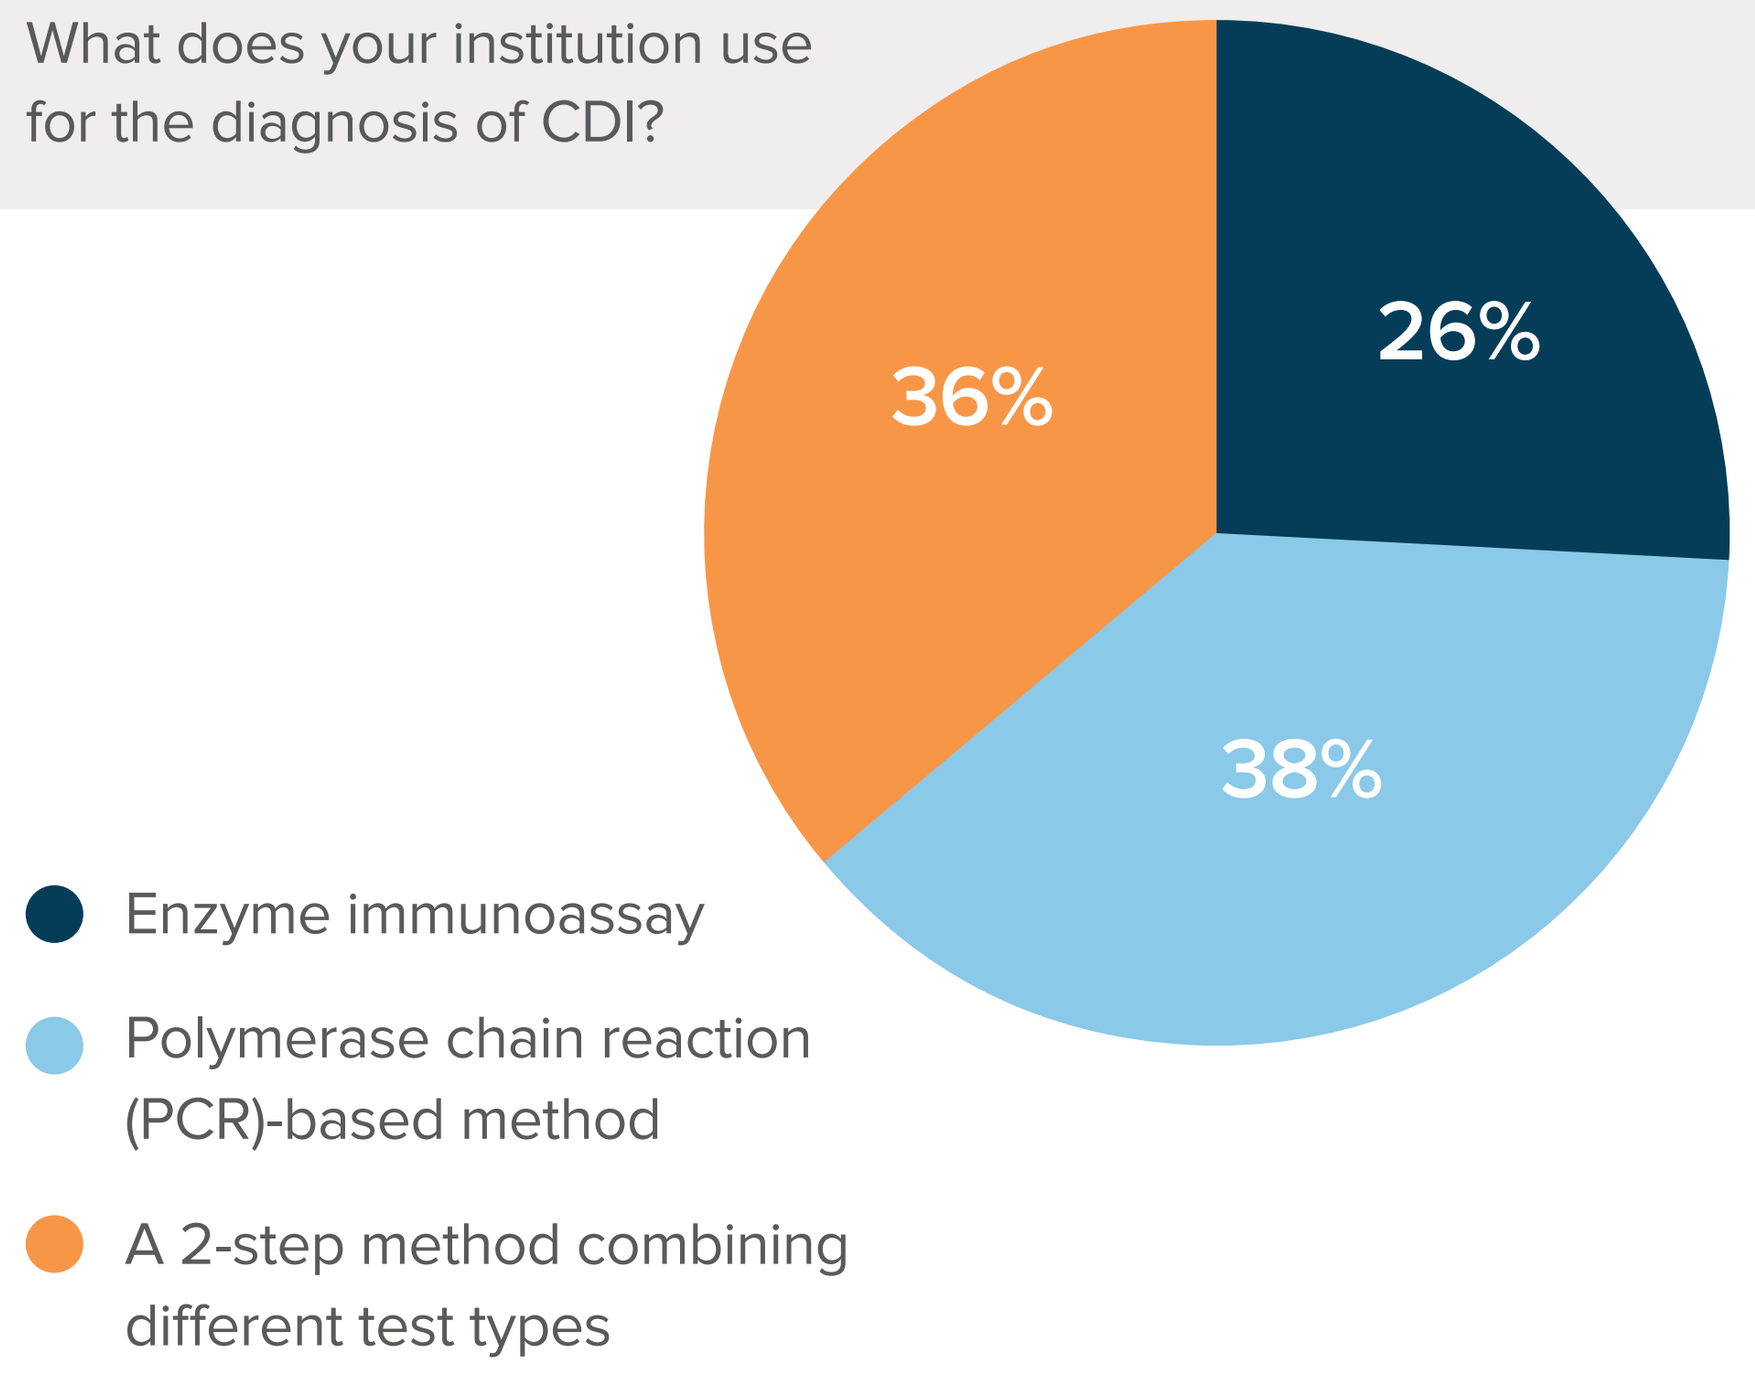
26%
38%
36%
Enzyme immunoassay
Polymerase chain reaction (PCR)-based method
A 2-step method combining different test types

NEW DEVELOPMENTS

About one-third (35%) are not aware of new strategies being investigated for prevention of CDI

Cephalosporins, penicillins, and carbapenems are beta-lactam antibiotics that are proven to damage gut microbiome diversity. Which of the following is a potential new strategy for reducing the negative effect of these medications on the gut microbiota?

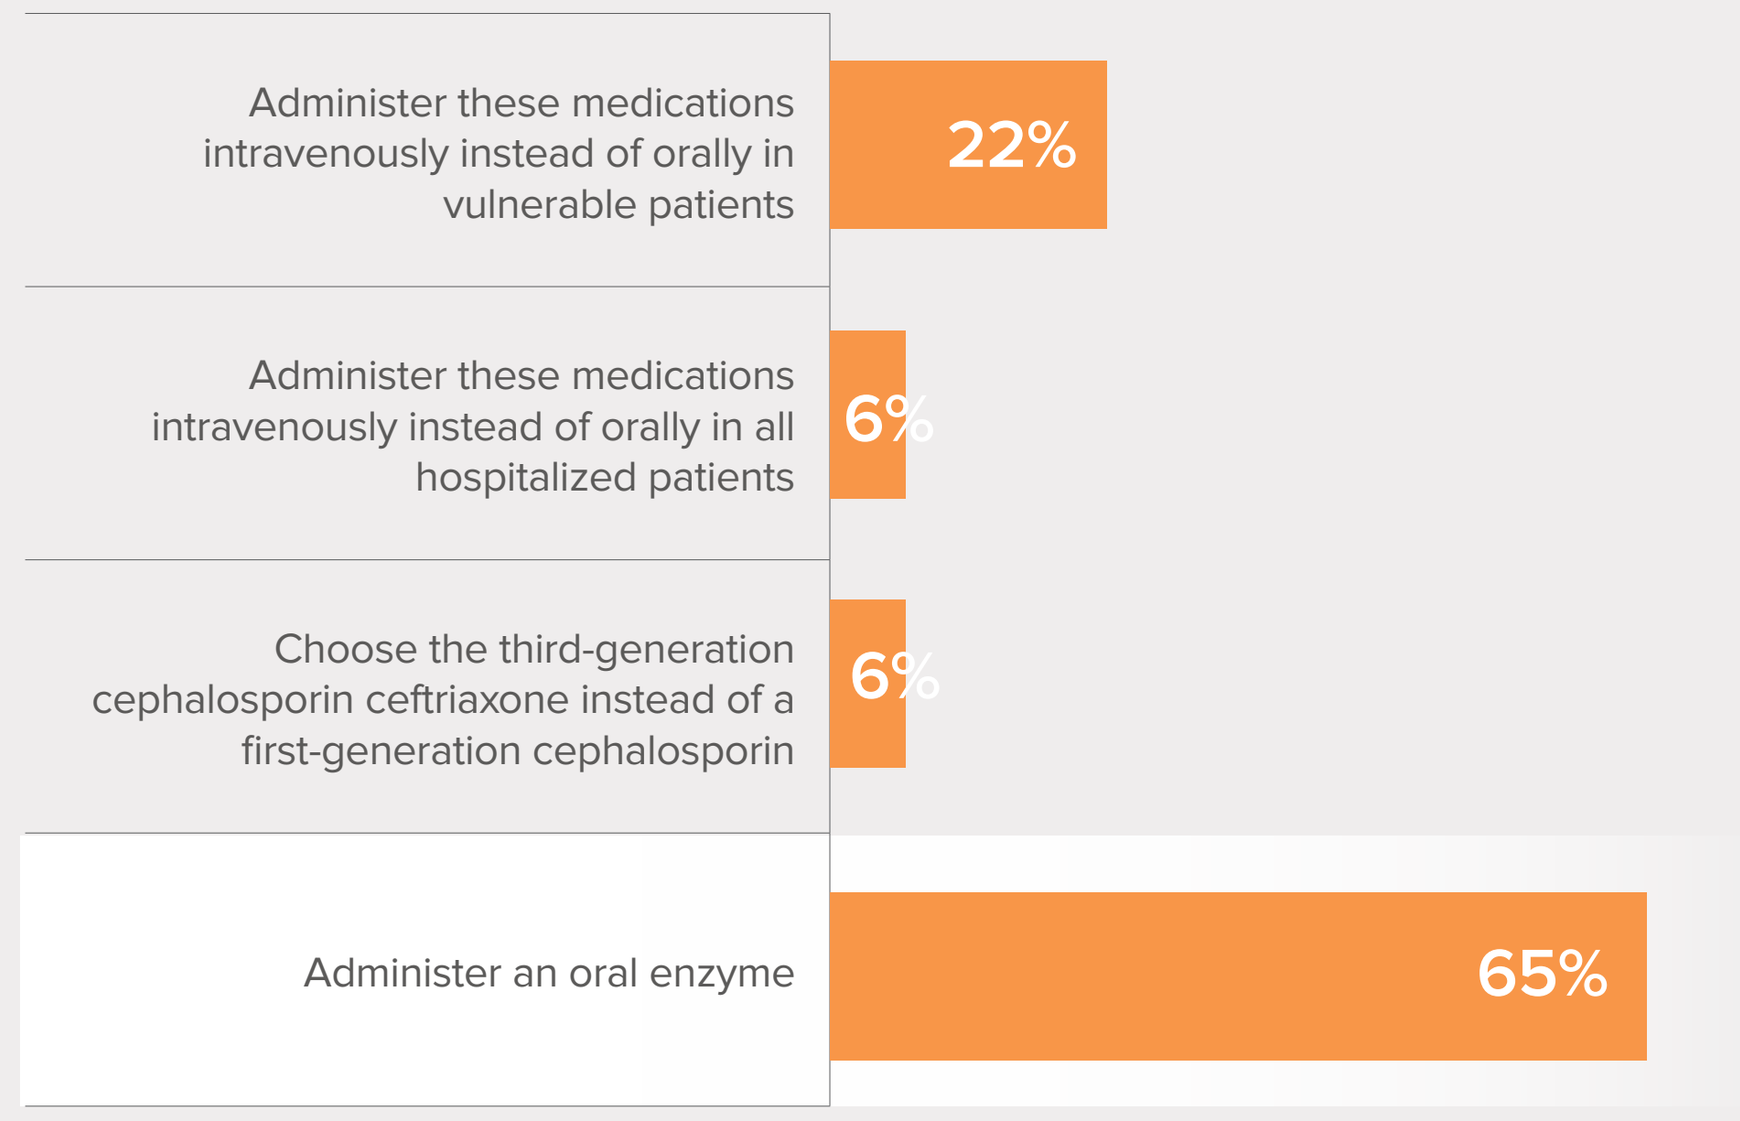
Administer these medications intravenously instead of orally in vulnerable patients
22%
Administer these medications intravenously instead of orally in all hospitalized patients
6%
Choose the third-generation cephalosporin ceftriaxone instead of a first-generation cephalosporin
6%
Administer an oral enzyme
65%

30% are not aware of the mechanism by which new therapies prevent development of CDI

What is the supposed mechanism by which ribaxamase, currently in clinical trials, may prevent CDI?

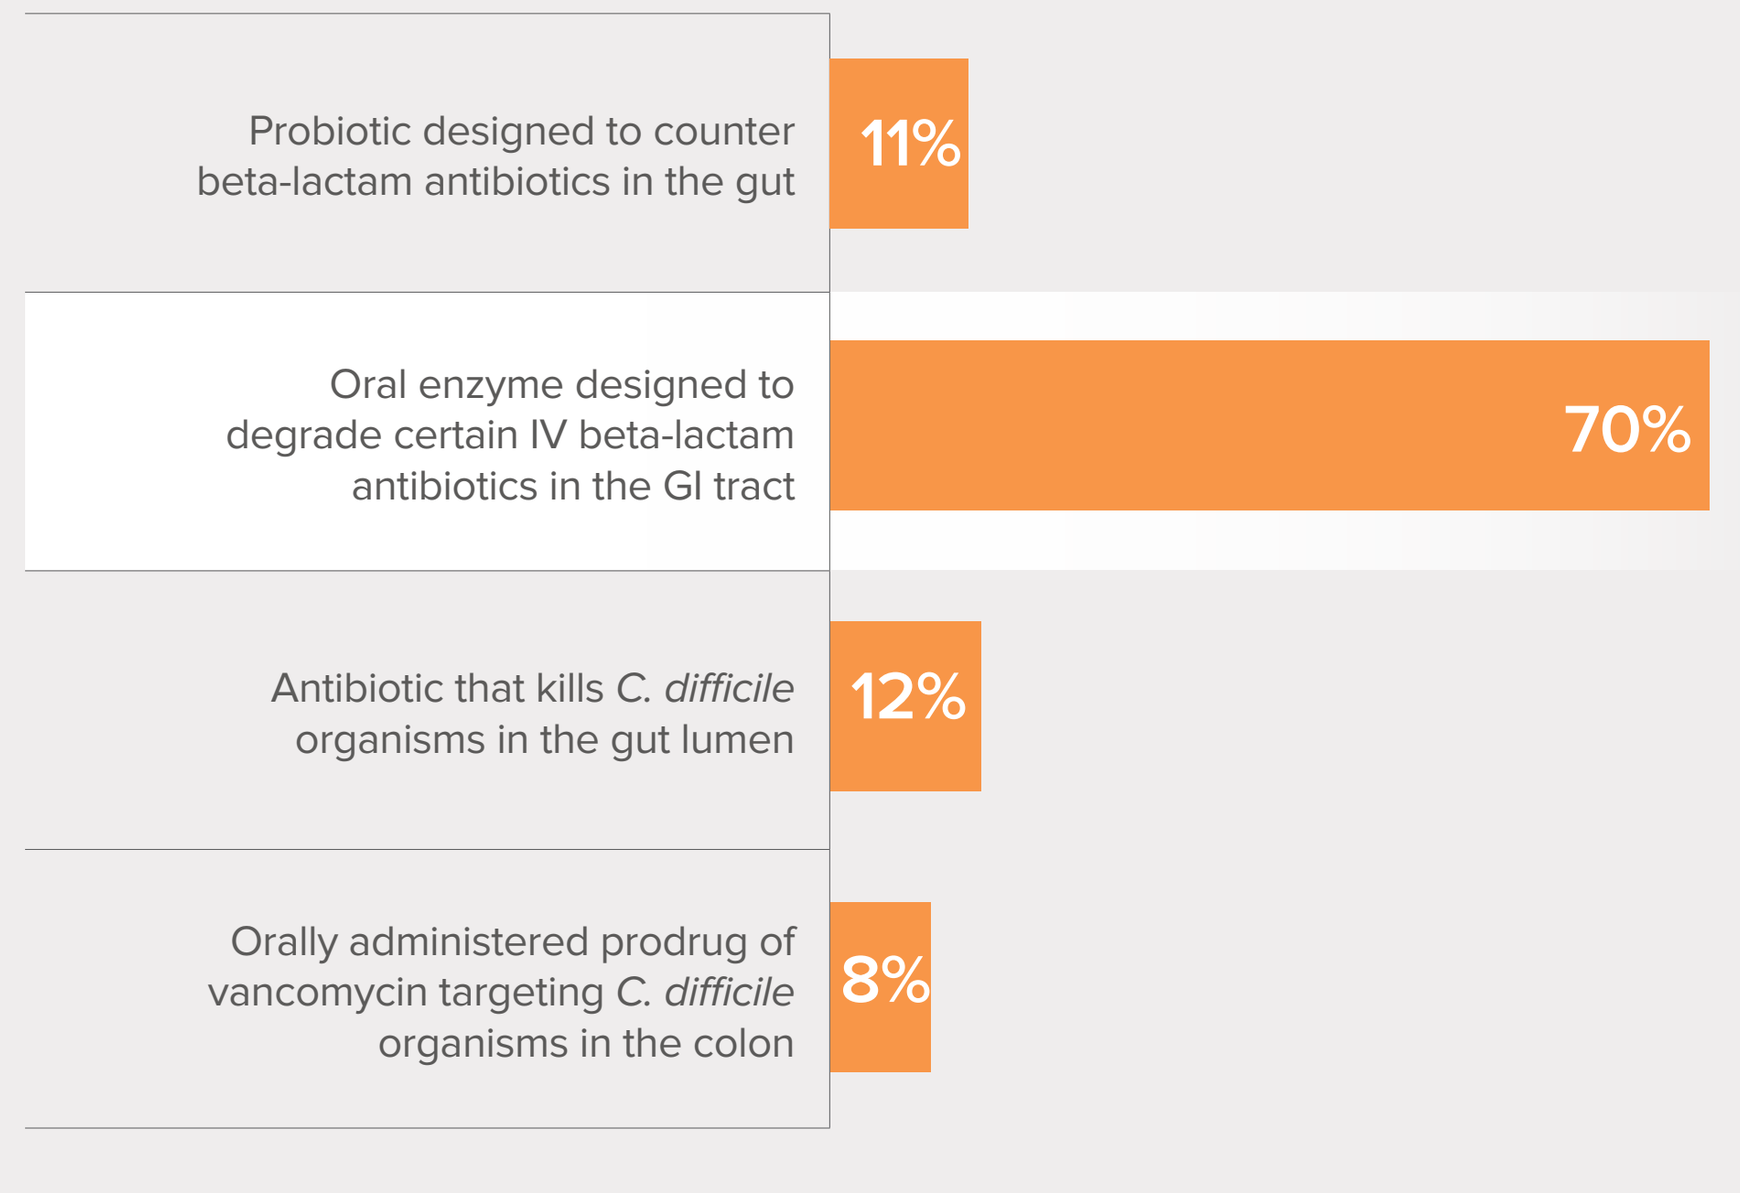
Probiotic designed to counter beta-lactam antibiotics in the gut
11%
Oral enzyme designed to degrade certain IV beta-lactam antibiotics in the GI tract
70%
Antibiotic that kills *C. difficile* organisms in the gut lumen
12%
Orally administered prodrug of vancomycin targeting *C. difficile* organisms in the colon
8%

CONCLUSION

This research yielded important insights into current clinical practice gaps among ID specialists regarding identification and prevention of CDI and could be valuable to inform needs for continuing medical education.

ACKNOWLEDGMENTS

This CME program was supported by an independent educational grant from Synthetic Biologics, Inc.

Scan here to view this poster online.