LBA11: PTX-35 Mediated Engagement of TNFRSF25 Induces an Activated and Immunosuppressive Treg Compartment Capable of Mitigating EAE Autoimmune Disease Severity

Charles J. Kroger[1], Matt Clark[1], Vikas Tahiliani[1], Eric P. Dixon[1], Rahul R. Jasuja[2], Matthew M. Seavey[1]

[1]Heat Biologics, Inc., Suite 400, 627 Davis Drive, Morrisville, NC, 27560 and [2]Pelican Therapeutics, 8122 Datapoint Drive, San Antonio, TX 78229

Introduction

CD4+ T cells expressing the transcription factor FOXP3 (Tregs) can prevent or suppress immune responses through both cell contact dependent and independent mechanisms. FoxP3+ Treg are critical mediators of peripheral tolerance. However, dysfunctional Treg development, fitness, or suppressive capacity negatively impacts peripheral tolerance and subsequently increases autoimmune disease susceptibility. Therapies targeting FoxP3+ Tregs are one potential strategy to treat autoimmune diseases by restoring peripheral tolerance. Tumor Necrosis Factor Receptor Superfamily member 25 (TNFRSF25) or Death Receptor 3 (DR3) is expressed at high levels on FoxP3+ Treg and has been shown to be a potent co-stimulatory molecule. DR3 agonism induced by the agonist antibody clone 4C12 induces significant Treg expansion *in vivo*, indicating a novel target for FoxP3+ Treg based immunotherapy. Through antibody engineering, 4C12 was humanized and affinity matured to recognize DR3 leading to the generation of PTX-35. Importantly both 4C12 and PTX-35 promote similar robust FoxP3+ Treg expansion, confirming DR3 mediated signaling was maintained following antibody modification. The goal of this study was to examine both the short- and long-term impact PTX-35 treatment has on FoxP3+ Treg fitness/function. Our results demonstrate PTX-35 engagement of DR3 activates and expands a FoxP3+ Treg population that has upregulated numerous immunosuppressive molecules and exhibit an increased immunosuppressive capability. Notably, PTX-35 treated mice exhibited minimal symptoms of MOG_{35-55} induced experimental autoimmune encephalomyelitis (EAE), which correlated with an increased FoxP3+ Treg compartment. Overall, these findings highlight the promising therapeutic potential of PTX-35 for the treatment of autoimmunity.

Methods

FoxP3+ Treg frequency and phenotypical experiments: C57BL/6 mice were injected *i.p.* with either PBS (day 0 control group), 10 mg/kg 4C12, or 10 mg/kg PTX-35. On the respective day post injection, either blood or splenic samples were analyzed for the frequency and/or phenotype of live CD4+ cells expressing FoxP3. Additionally, the expression level of DR3 on different T cell populations directly *ex vivo* and on the binding of PTX-35 on day 7 using anti-mouse IgG1. For the Treg suppression assay, C57BL/6-FoxP3EGFP mice were injected with either PBS or 10 mg/kg PTX-35. On day 5 post injection, CD4+FoxP3EGFP+ cells were sorted then co-cultured at different ratios with CTV labeled CD4+ splenocytes for 72 hrs. The percent of proliferating CTV CD4+ Teff cells was determined by flow cytometry. Samples were acquired on a Beckman Coulter Cytoflex S instrument and analyzed using FlowJo.

EAE experiment: C57BL/6 mice were immunized with the MOG_{35-55} Kit manufactured by Hooke Laboratories. Day 12 and 16 post immunization a group of mice were treated with 10 mg/kg PTX-35. Another cohort of mice were injected with an isotype control antibody at 10 mg/kg. Mice were regularly monitored for weight loss and paralytic score (paralytic score index: 0 = no paralysis, 1 = limp tail, 2 = limp tail + one hind limb weakness, 3 = limp tail + complete hind leg paralysis, 4 = limp tail, complete hind leg paralysis, and front limb weakness). At the end of the study, the spinal cord was harvested for histological analysis and the frequency of CD4+ T cells expressing FoxP3+ was examined by flow cytometry

Unpaired t-test statistical analysis: *p<.05, **p<.01, ***p<.001, ****p.0001 was completed using GraphPad Prism software.

Figures

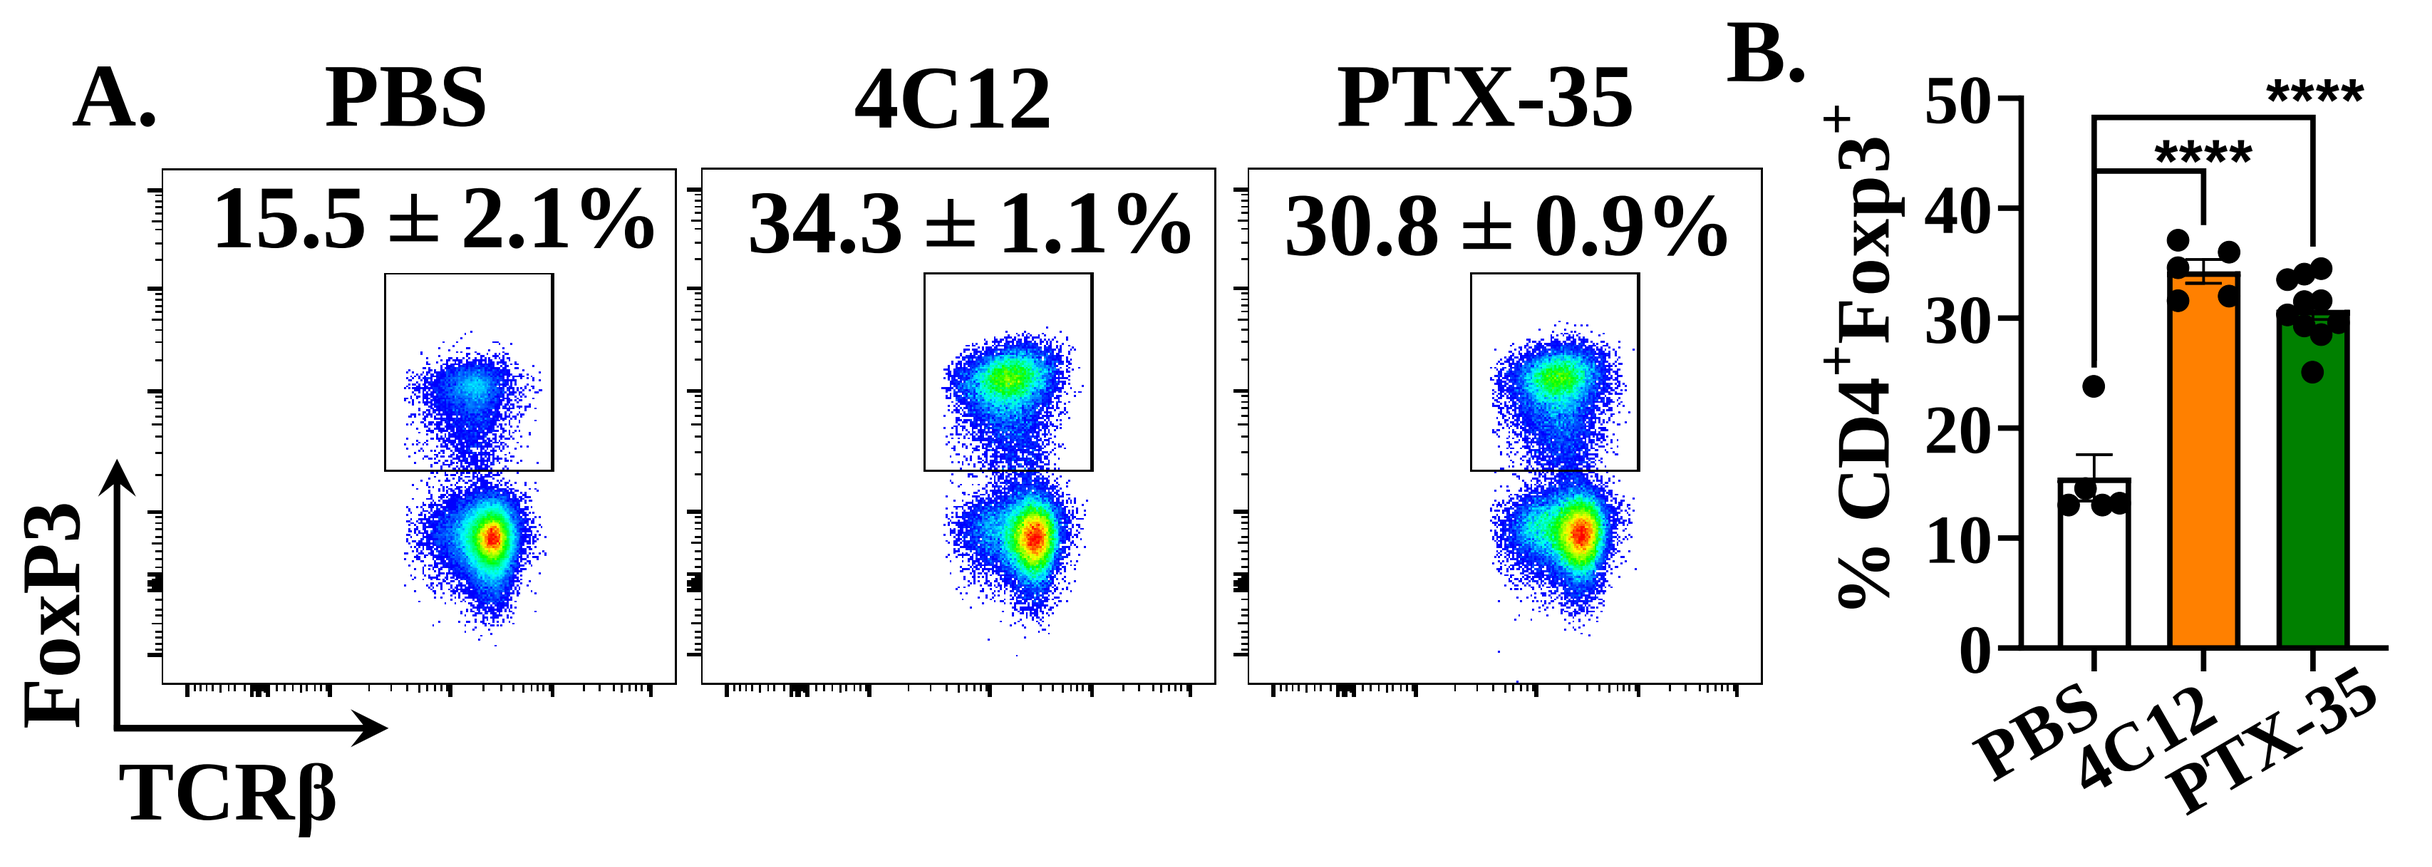
A. PBS 4C12 PTX-35
15.5 ± 2.1%
34.3 ± 1.1%
30.8 ± 0.9%
FoxP3
TCRβ
B.
% CD4+Foxp3+
PBS 4C12 PTX-35

Figure 1. PTX-35 engagement of DR3 promotes a similar increased peripheral Treg compartment compared to its parental antibody clone 4C12. (A) The frequency of splenic CD4+ T cells expressing FoxP3 was examined on day 4-5. The mean ± SEM is included in the respective histograms. (B) The results from two independent experiments pooled together (n≥5 mice/group).

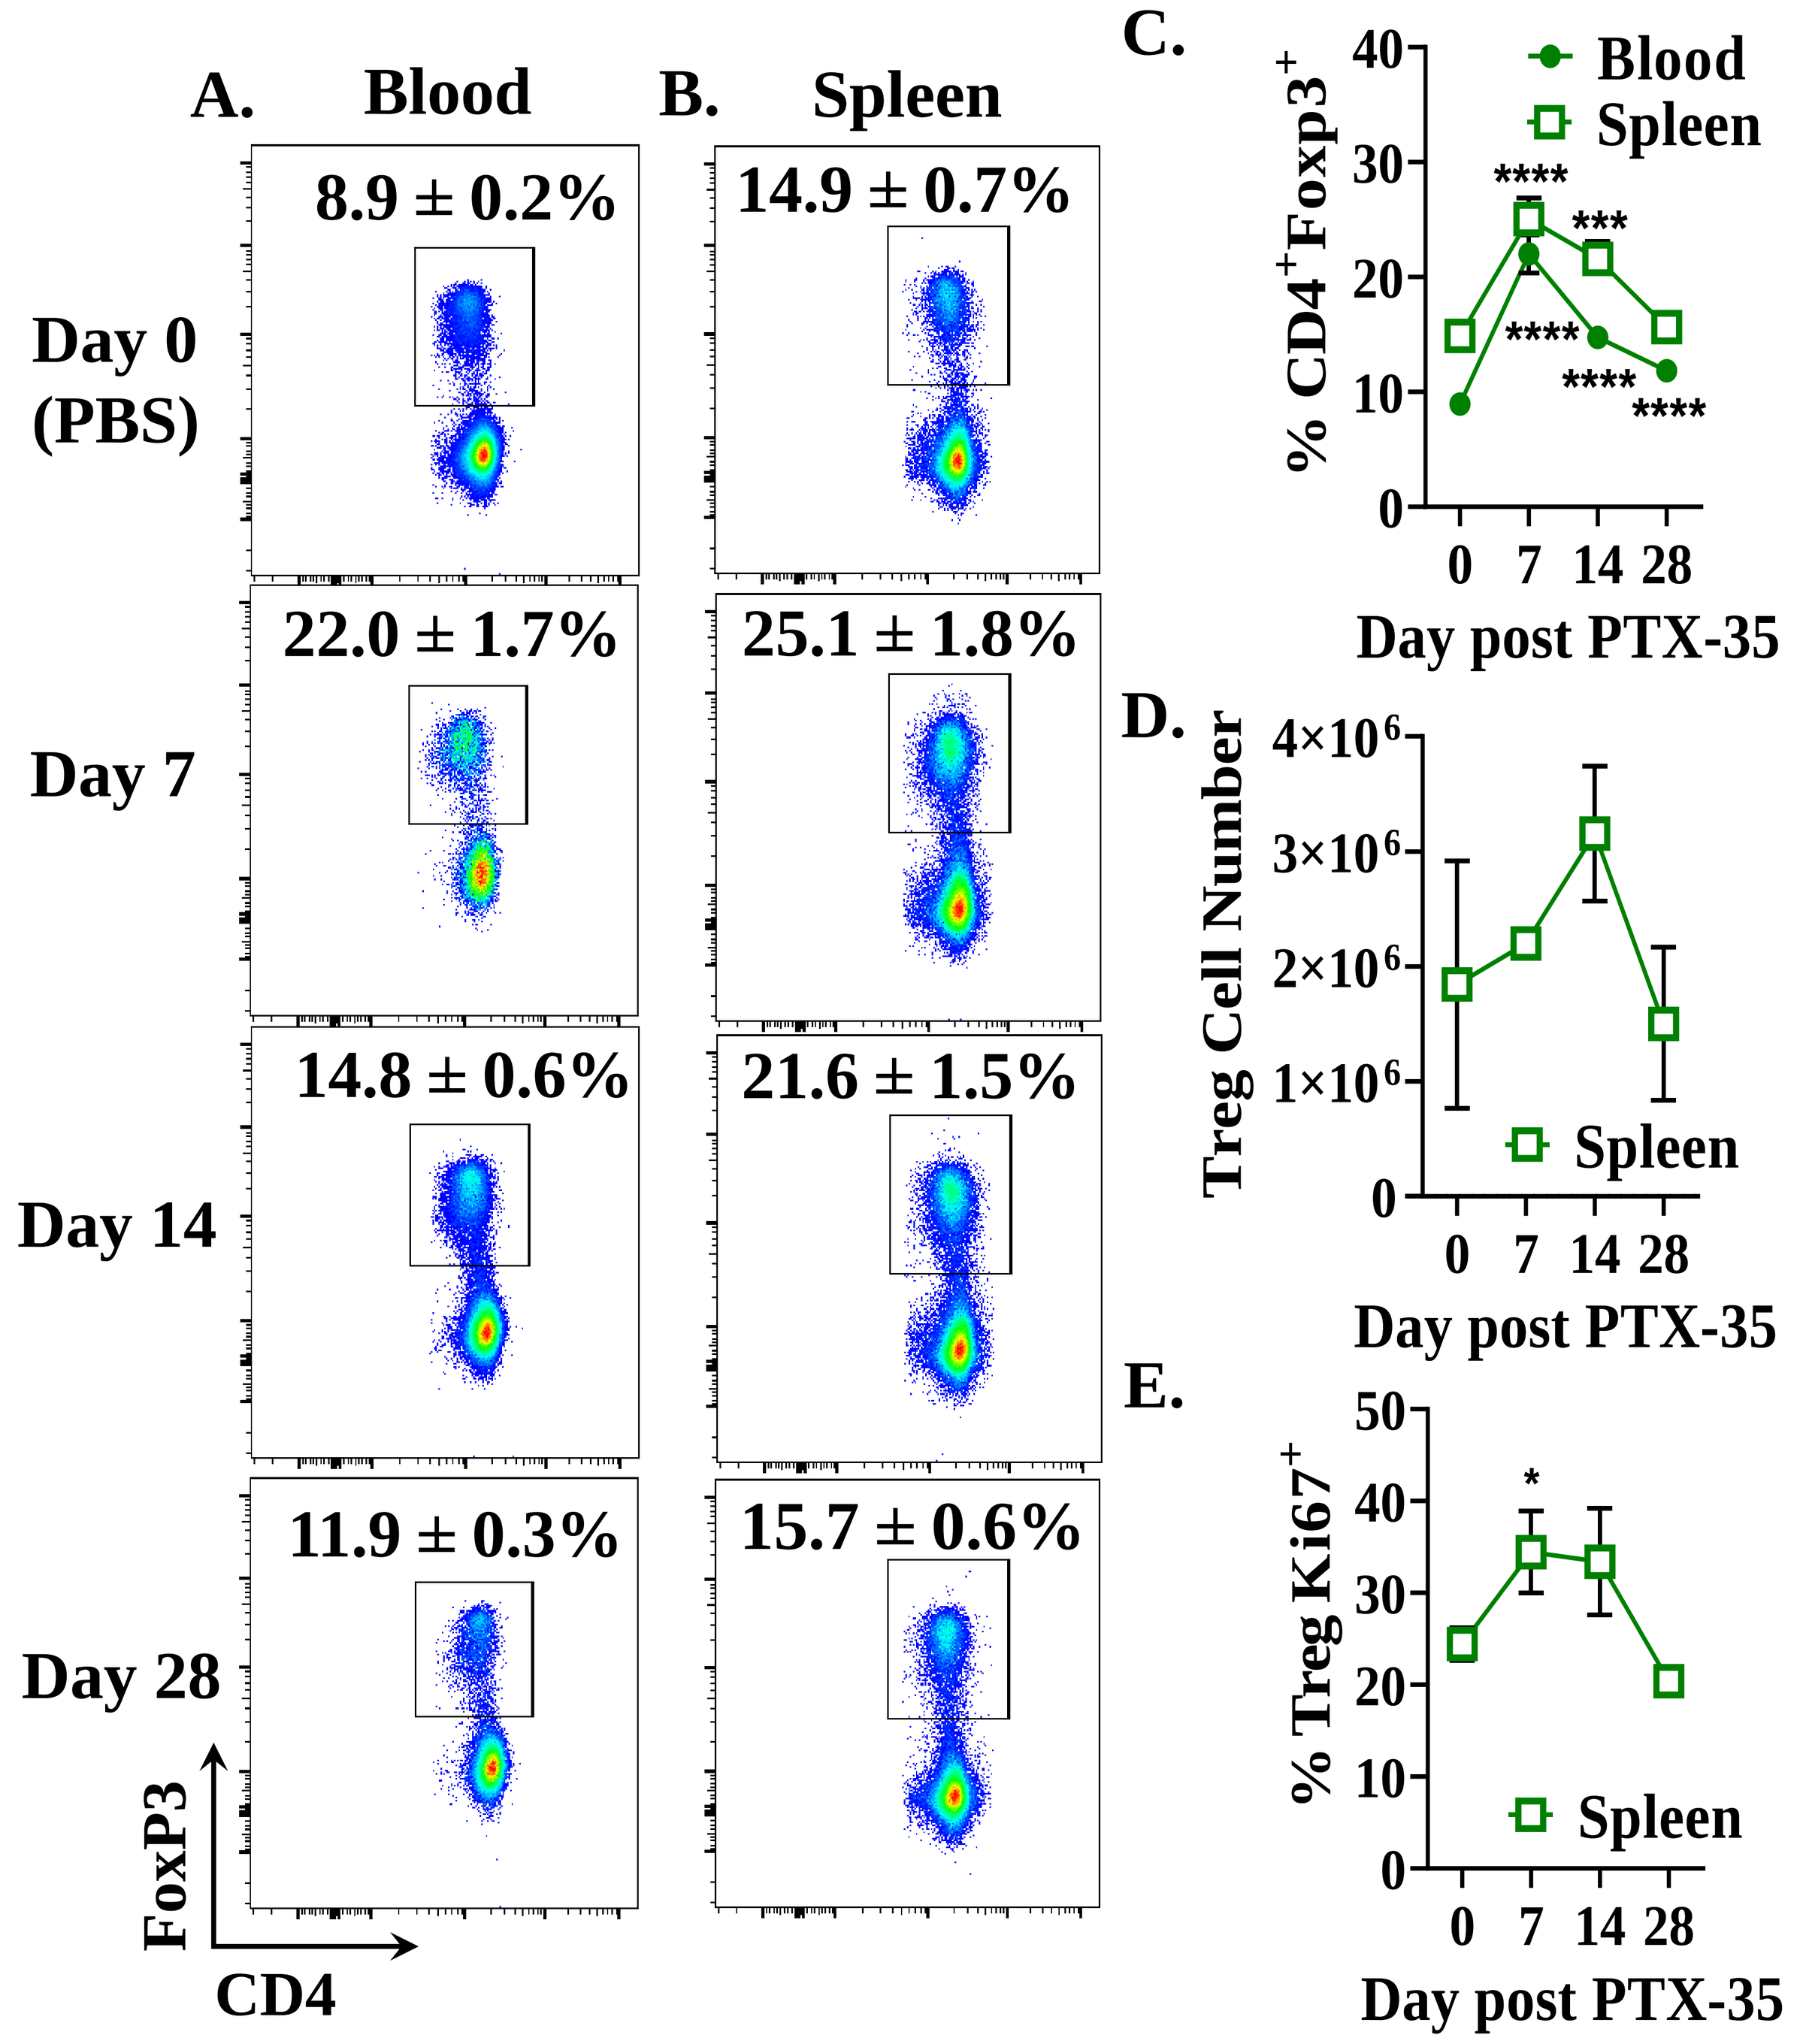
A. Blood
B. Spleen
Day 0 (PBS)
8.9 ± 0.2%
14.9 ± 0.7%
Day 7
22.0 ± 1.7%
25.1 ± 1.8%
Day 14
14.8 ± 0.6%
21.6 ± 1.5%
Day 28
11.9 ± 0.3%
15.7 ± 0.6%
FoxP3
CD4
C. % CD4+Foxp3+
Blood
Spleen
Day post PTX-35
D. Treg Cell Number
Spleen
Day post PTX-35
E. % Treg Ki67+
Spleen
Day post PTX-35

Figure 2. A single dose of PTX-35 induces significant Treg expansion that persists for weeks. The frequency of CD4+ T cells expressing FoxP3 in the blood (A,C) and spleen (B,C) was examined by flow cytometry on day 0 (PBS), 7, 14, or 28. The mean ± SEM is included in the respective histograms. (C) Pooled results from two independent experiments analyzing the Treg frequency in the blood and spleen (n≥8 mice/group). (D) The number of FoxP3+ Tregs in the spleen was calculated (n=2-4 mice/group). (F) The proliferation profile of CD4+FoxP3+ T cells was investigated by staining for Ki67 (n≥8 mice/group).

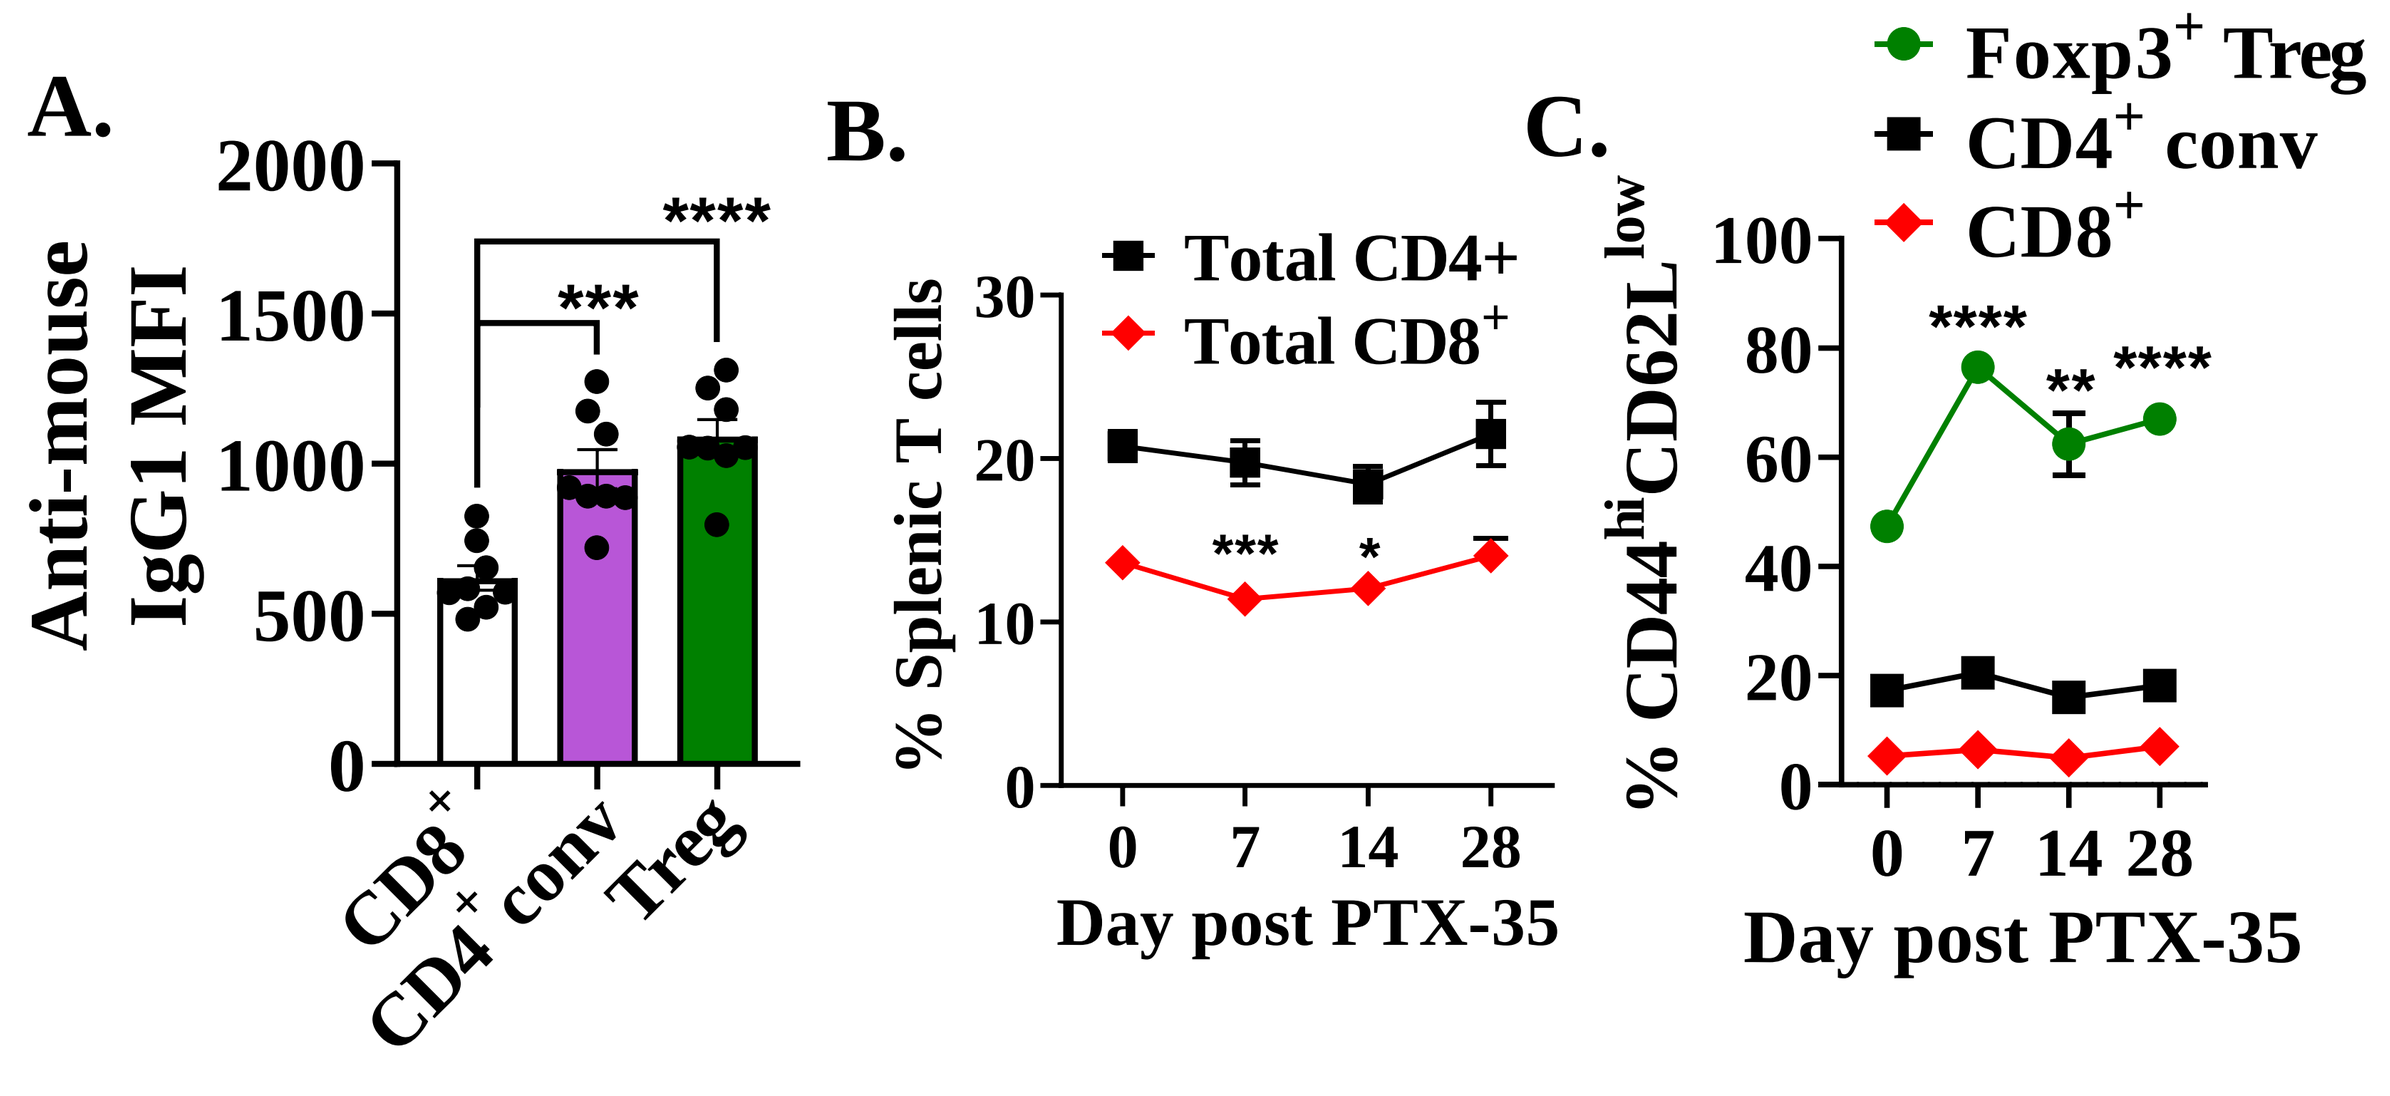
A. Anti-mouse IgG1 MFI
CD8+ CD4+ conv Treg
B. % Splenic T cells
Total CD4+
Total CD8+
Day post PTX-35
C. % CD44hiCD62Llow
Foxp3+ Treg
CD4+ conv
CD8+
Day post PTX-35

Figure 3. PTX-35 can bind to but does not expand or activate the conventional splenic T cell populations in healthy mice. (A) The binding of PTX-35 on different splenic T cell populations was investigated on day 7.(B) The total frequency of CD8+ and CD4+ T cells was examined in the spleen on day 7. (C) The activation status of FoxP3+ Treg, CD4+ conventional, and CD8+ T cells was examined temporally in the spleen after PTX-35 injection (n≥8 mice/group).

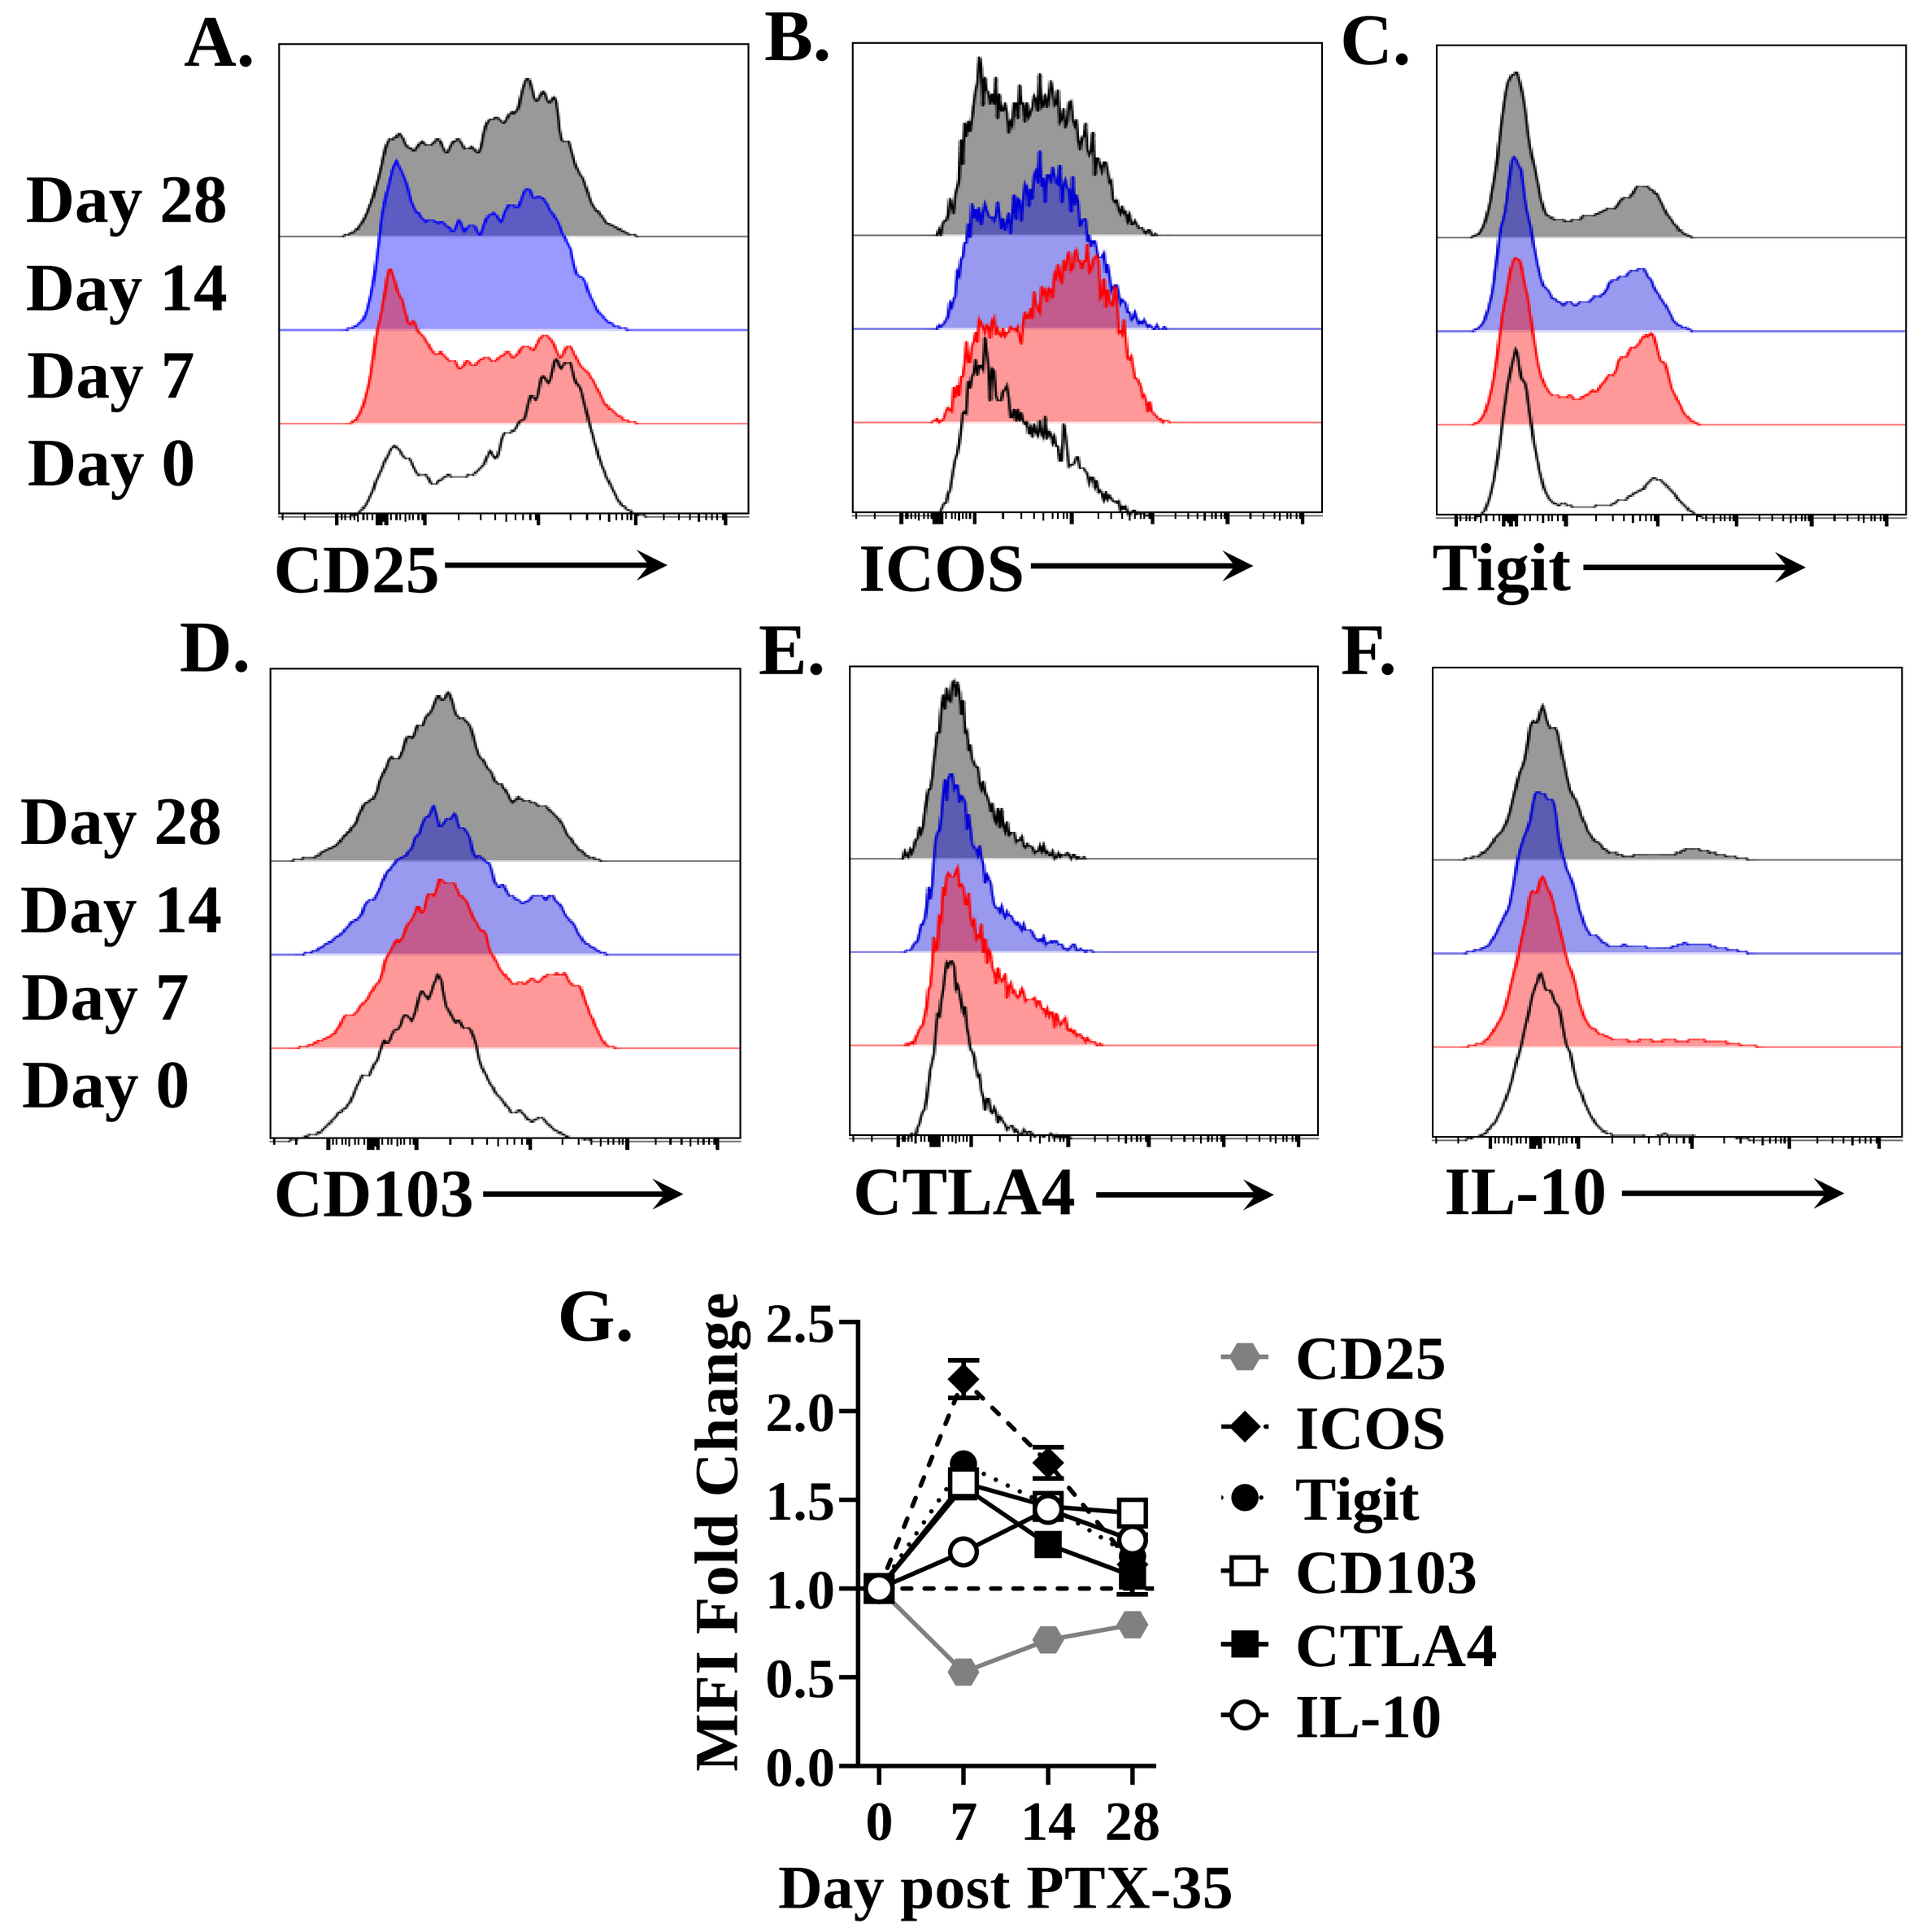
Day 28
Day 14
Day 7
Day 0
CD25
ICOS
Tigit
CD103
CTLA4
IL-10
G. MFI Fold Change
Day post PTX-35

Figure 4. A single dose of PTX-35 induces significant Treg activation and upregulation of numerous immuno-suppressive associated markers that endures for weeks. (A-G) The expression profile of immunosuppressive associated molecules on splenic FoxP3+ Tregs was determined at the indicated time points following PTX-35 treatment: (A) CD25, (B) ICOS, (C) Tigit, (D) CD103, (E) CTLA4, (F) IL-10. (G) The MFI fold change for the respective molecules was calculated over the PBS treated mouse control levels (n≥2 mice/time point).

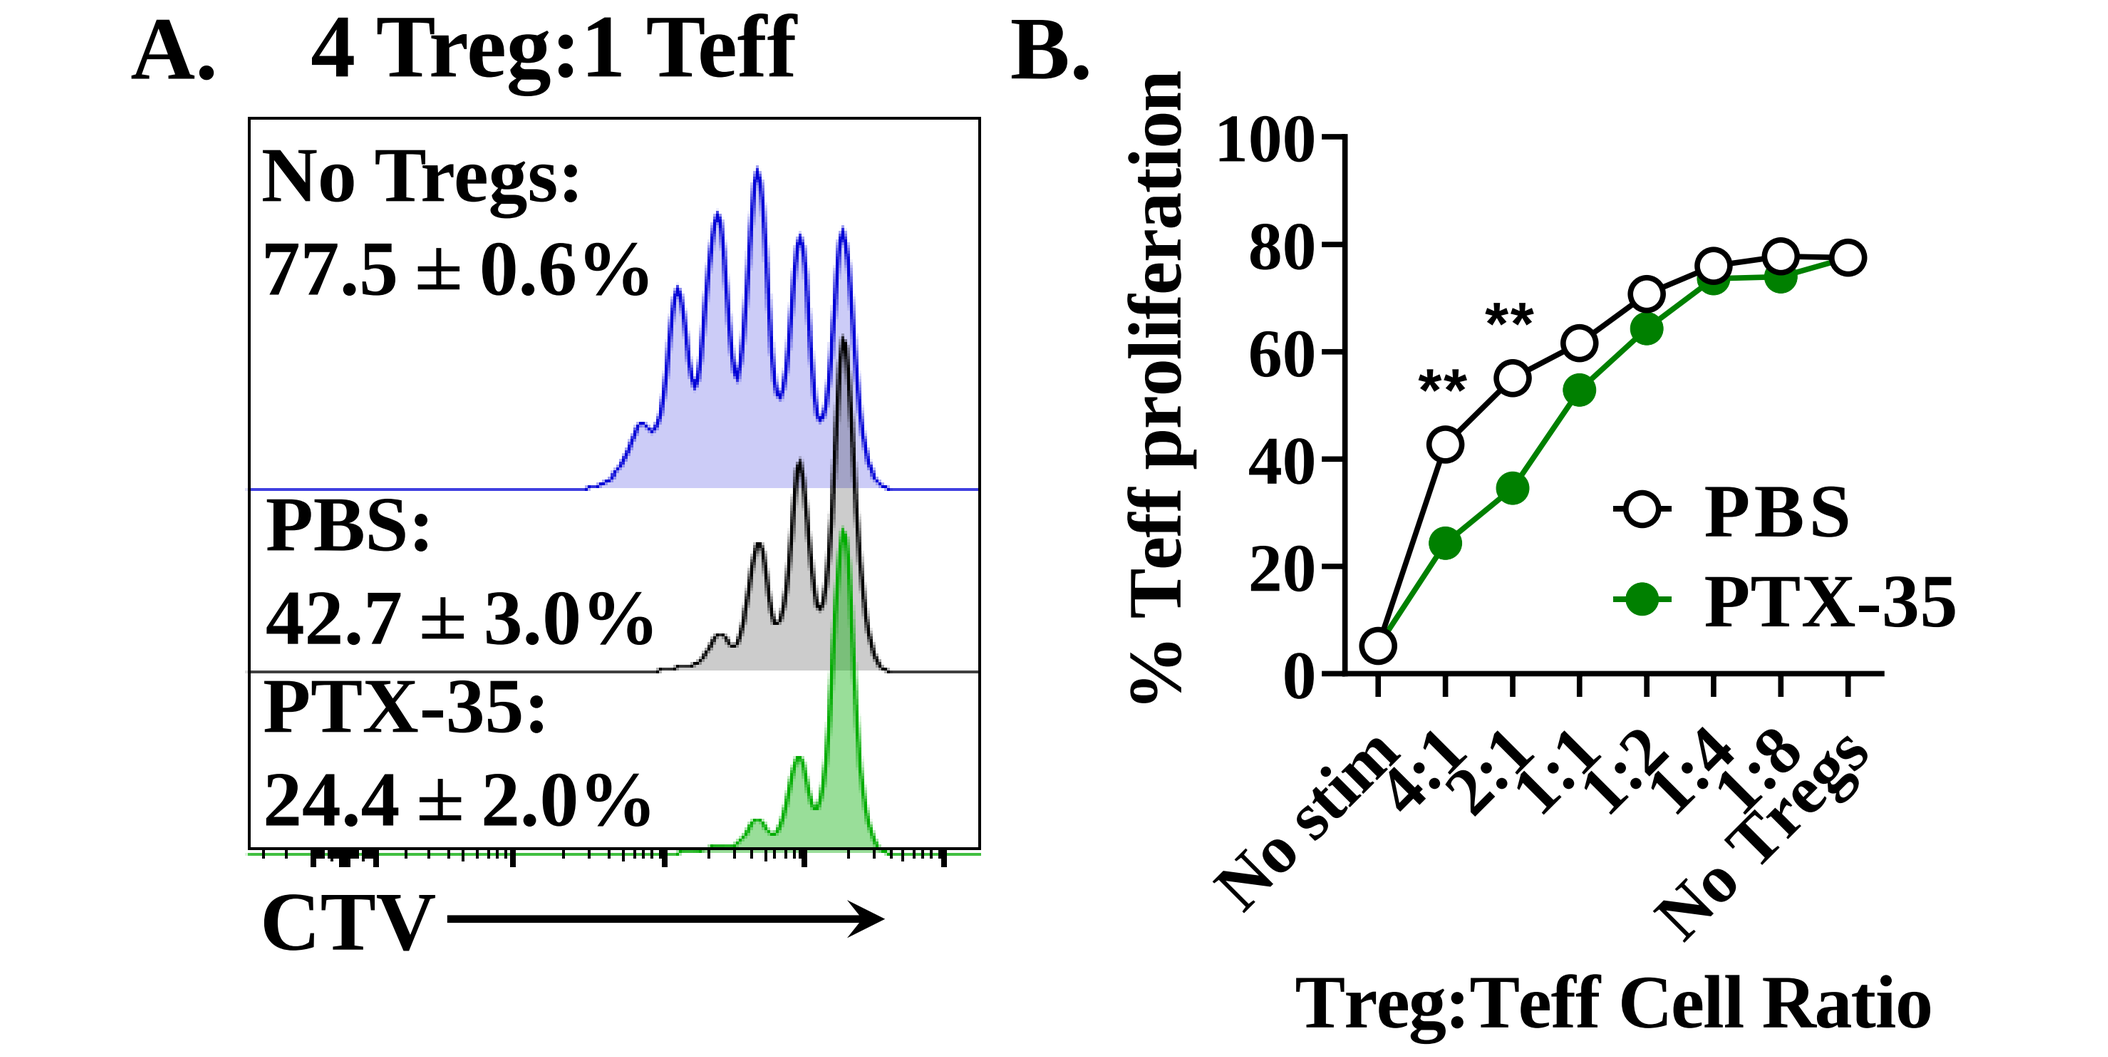
A. 4 Treg:1 Teff
No Tregs: 77.5 ± 0.6%
PBS: 42.7 ± 3.0%
PTX-35: 24.4 ± 2.0%
CTV
B. % Teff proliferation
PBS
PTX-35
No stim 4:1 2:1 1:1 1:2 1:4 1:8 No Tregs
Treg:Teff Cell Ratio

Figure 5. Tregs enriched from PTX-35 treated mice exhibit enhanced suppressive capacity. (A) Sorted FoxP3EGFP+ Tregs were cultured at 4:1 ratio with with CTV labeled CD4+ T cells (Teff) then proliferation examined by flow cytometry 72 hrs later. (B) The Teff proliferation profile at numerous cell ratios after isolating FoxP3EGFP+ cells from multiple mice (n≥4 mice).

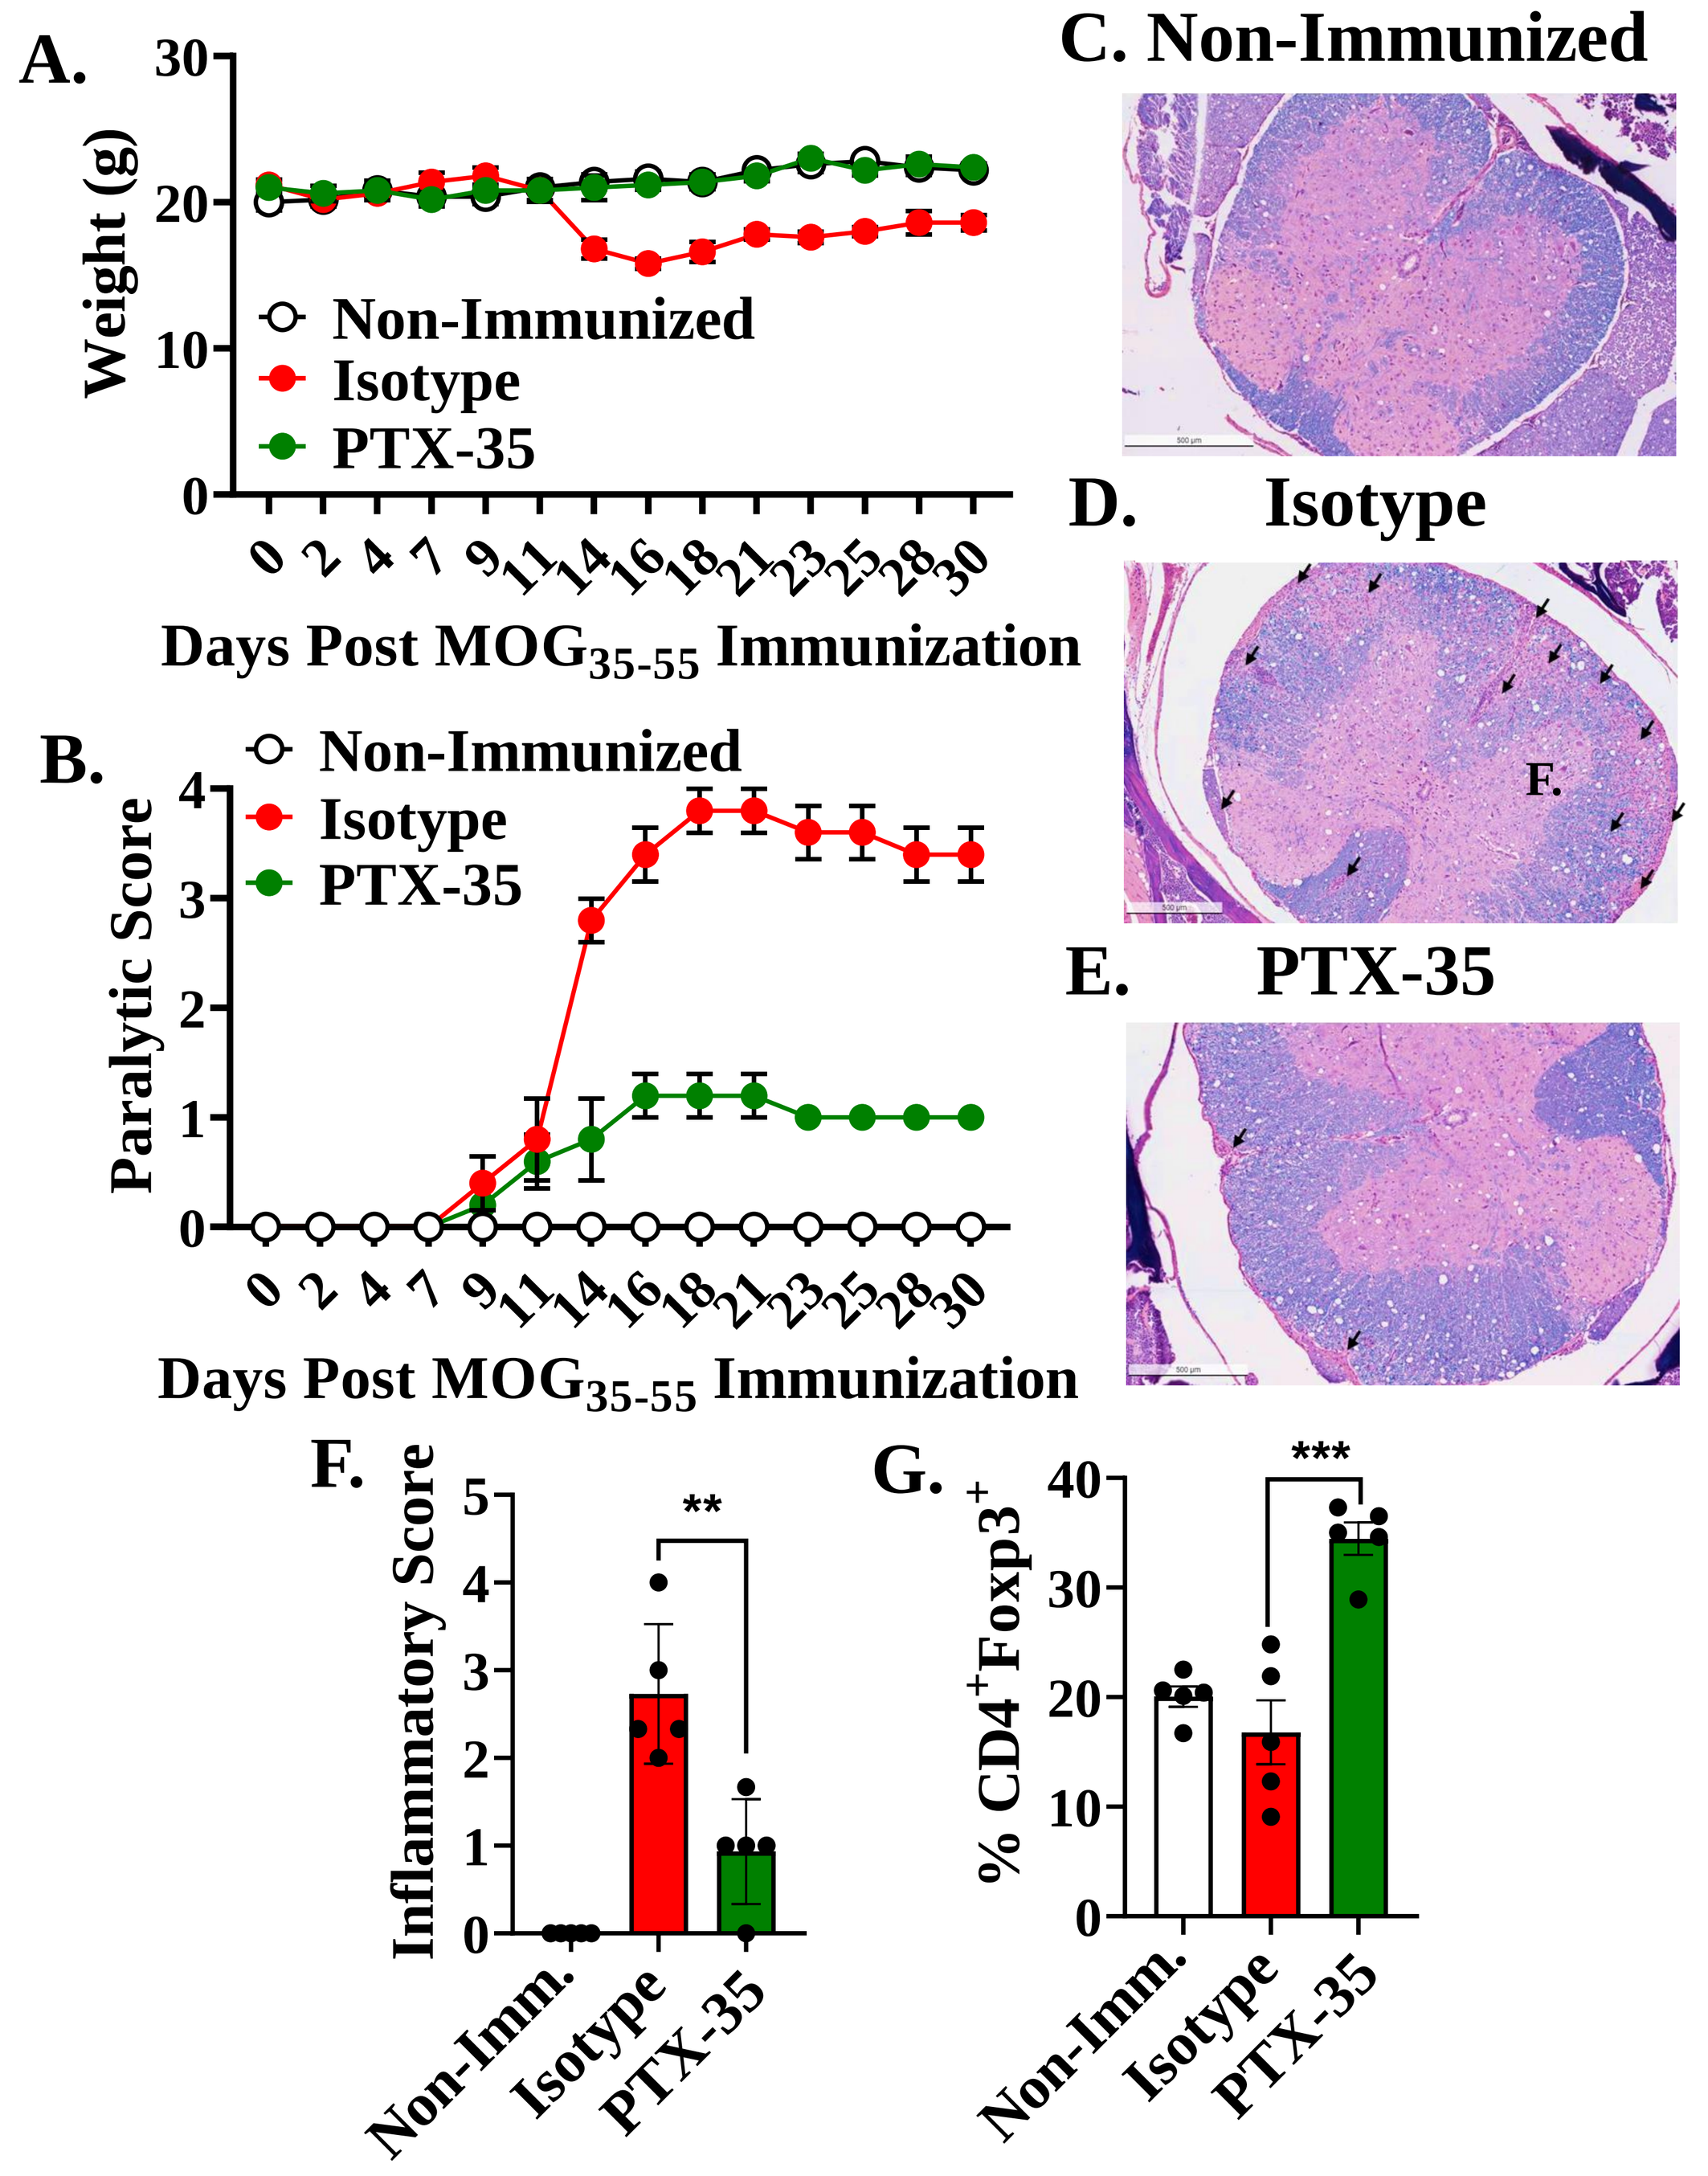
A. Weight (g)
Non-Immunized
Isotype
PTX-35
Days Post MOG35-55 Immunization
B. Paralytic Score
Non-Immunized
Isotype
PTX-35
Days Post MOG35-55 Immunization
C. Non-Immunized
D. Isotype
E. PTX-35
F. Inflammatory Score
Non-Imm. Isotype PTX-35
G. % CD4+Foxp3+
Non-Imm. Isotype PTX-35

Figure 6. PTX-35 treatment reduces EAE disease severity and corresponds with an increased frequency of FoxP3+ Treg. The (A) weight and (B) paralytic score were recorded after MOG immunization. (C-E) Luxol Fast Blue staining of spinal cord sections from (C) Non-Immunized, (D) Isotype, or (E) PTX-35 treated mice. The black arrows highlight lymphocytic infiltration. (F) Histological scoring of spinal cord tissue inflammation from multiple mice. (G) The frequency of splenic CD4+ T cells expressing FoxP3 at the end of the study (n=5 mice).

Conclusions

PTX-35 is a potent DR3/TNFRSF25 agonist antibody that enhances the peripheral tolerance potential of treated mice by:

- Increasing the frequency and number of circulating and tissue resident FoxP3+ Tregs by promoting cellular proliferation
- Selectively activating the FoxP3+ Treg population, with only transient effects on CD8+ T cells
- Inducing upregulation of numerous cell contact dependent and independent immunosuppressive molecules for an extended time period
- Increasing the overall suppressive capacity of FoxP3+ Treg dampening Teff proliferation

This altered FoxP3+ Treg compartment reduced disease pathology in the EAE autoimmune mouse model, which correlated with an elevated peripheral FoxP3+ compartment

Model

Circulating peripheral FoxP3+ Treg express high basal levels of DR3, that following engagement with PTX-35 promotes increased Signal 2 events (costimulatory) driving FoxP3+ Treg activation, expansion, and upregulation of immunosuppressive molecules.

The modified FoxP3+ Treg population can migrate into inflamed tissue and more effectively dampen the ongoing autoimmune response via both cell contact dependent and independent mechanisms, generating an anti-inflammatory environment, restoring peripheral tolerance, and preserving tissue health and function.

Acknowledgements

We appreciate the effort of Dr. Ramon Coronado, Pelican Therapeutics CEO, for critical review of the PTX-35 data. We also would like to thank Dr. Robert Levy at University of Miami for informative discussions that have shaped the research project. Additionally, we would also like to acknowledge the contributions of former Heat Biologics employee Taylor Schreiber and current employees Abra Chaney and Anh Trinh.

*Corresponding Author: Charles Kroger
ckroger@heatbio.com

References

1. Schreiber T.H. et al. Therapeutic Treg expansion in mice by TNFRSF25 prevents allergic lung inflammation. J. Clin. Inv. 2010, 120:3629-3640.
2. Schreiber T.H. et al. Immunobiology of TNFSF15 and TNFRSF25. Immunol Res. 2013, 57:3-11.
3. Nishikii H. et al. DR3 signaling modulates the function of Foxp3+ regulatory T cells and the severity of acute graft-versus-host disease. 2016. Blood. 128:2846-2858.
4. Yu Y. et al. Analysis of therapeutic potential of preclinical models based on DR3/TL1A pathway modulation. Exp. and Therap. Med. 2021. DOI://10.3892/etm.2021.10125.