Induction of KLF4 by LOR-253 as an innovative therapeutic approach to induce apoptosis in acute myeloid leukemia.

Ronnie Lum, Mojib Javadi, Tiffany Cheng, Robert Peralta, Howard Cukier, Jeff Lightfoot, Yoon Lee, Aiping Young, and William G. Rice
Lorus Therapeutics Inc., Toronto, Ontario, Canada

4544

Abstract

LOR-253, a small molecule anticancer agent that acts through induction of the tumor suppressor Krüppel-like factor 4 (KLF4), has demonstrated antitumor activity as a single agent in a Phase I study in patients with advanced or metastatic solid tumors. Recently, the vast majority of patients with acute myeloid leukemia (AML) were shown to inappropriately express the embryonic CDX2 gene in bone marrow stem and progenitor cells, resulting in down-regulation of KLF4 expression, speculated to be the leukemogenic event. Consequently, we examined the antitumor activity and mechanism of action for LOR-253 in AML cells and cells representing other hematological malignancies. Indeed, LOR-253 was found to inhibit proliferation of various human leukemia and lymphoma cell lines in vitro with low nM IC_{50} values. Furthermore, LOR-253 induced high levels of KLF4 mRNA expression in AML cells and a resultant significant increase in expression of p21, a cyclin-dependent kinase inhibitor that is transcriptionally regulated by KLF4. Consistent with these findings, in AML cells we found that LOR-253 induced G1/S cell cycle arrest and apoptosis, based on positive Annexin V staining, activated caspase-3, and increased BAX mRNA expression. Studies are underway to further characterize the pathway that mediates KLF4 induction by LOR-253, to characterize the effects of LOR-253 in combination with approved chemotherapies for AML, and to assess the efficacy of LOR-253 in animal models of AML. Induction of KLF4 represents a novel approach to the treatment of AML and other hematologic malignancies, and LOR-253 is the only clinical stage agent to act through this mechanism of action.

Introduction

LOR-253 is a novel small molecule being developed by Lorus Therapeutics as an anticancer agent for treatment of hematologic cancers. In preclinical studies, LOR-253 has shown significant anticancer activity in a range of tumor types, including non-small cell lung cancer (NSCLC) and colon cancer, with minimal toxicity at efficacious doses. A Phase 1 study with LOR-253 was recently completed in patients with advanced or metastatic tumors, demonstrating evidence of antitumor activity with minimal toxicity.

Mechanism of action and efficacy studies have revealed that the anticancer activity of LOR-253 is associated with induction of expression of KLF4, a tumor suppressor that is downregulated in several cancers including colon and NSCLC. Several reports have shown that KLF4 expression is regulated by the homeobox gene CDX2 in colon cancer and leukemia. Notably, the vast majority of patients with acute myeloid leukemia (AML) were shown to aberrantly express CDX2 in bone marrow stem and progenitor cells, resulting in downregulation of KLF4 expression as the potential leukemogenic event (Scholl et al., 2007). Cdx2 is highly leukemogenic in experimental models of AML, in that mice transplanted with bone marrow cells expressing Cdx2 rapidly develop fatal AML (Rawat et al., 2008). CDX2 was recently shown to act as a transcriptional repressor of KLF4 expression in leukemia by binding to CDX2 consensus sequences within the KLF4 promoter. Once bound, CDX2 recruits the H3K4 demethylase KDM5B and silences KLF4 transcription by an epigenetic mechanism (Faber et al., 2013). These observations suggest that CDX2 promotes leukemogenesis by suppression of KLF4.

LOR-253 is currently the only investigational therapy that specifically induces expression of KLF4 in cancer. In the current study, we show that LOR-253 is highly active in preclinical AML models, and that KLF4 induction by LOR-253 correlates with anticancer activity in AML.

CDX2/KLF4 Expression in AML

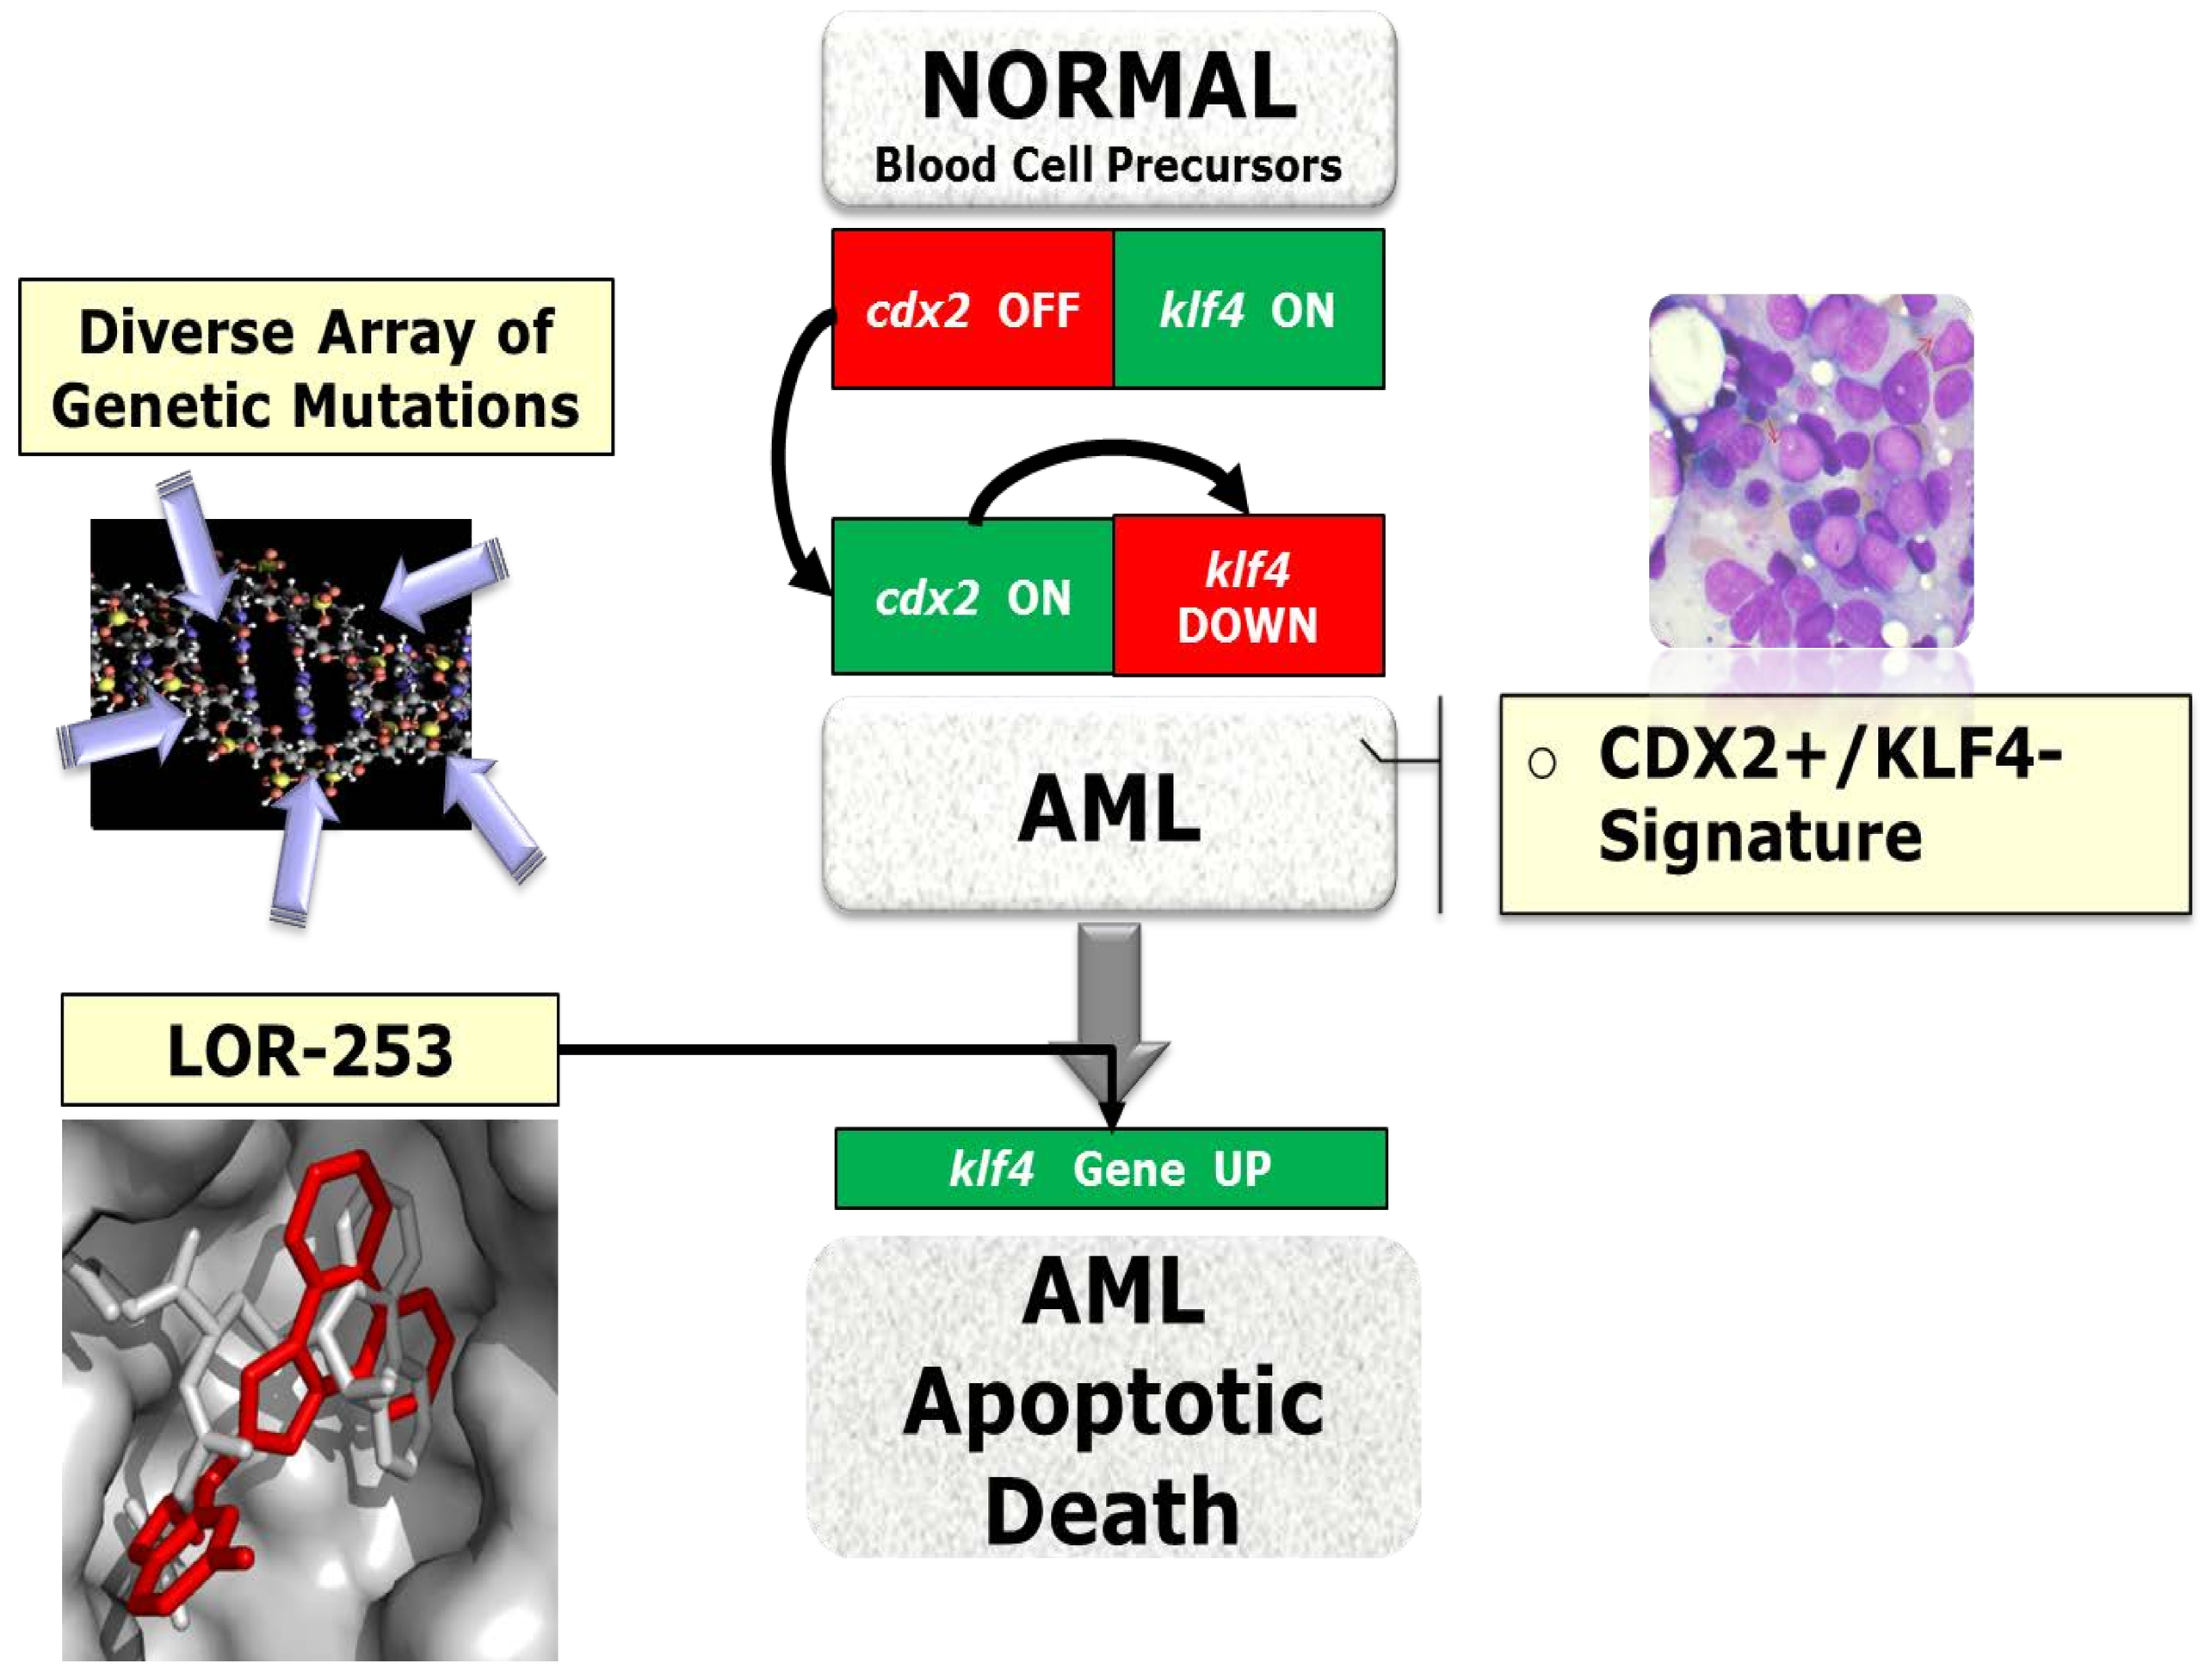
NORMAL
Blood Cell Precursors
cdx2 OFF
klf4 ON
Diverse Array of Genetic Mutations
cdx2 ON
klf4 DOWN
AML
CDX2+/KLF4- Signature
LOR-253
klf4 Gene UP
AML Apoptotic Death

Figure 1. CDX2 is a homeobox transcription factor that functions in embryonic organogenesis and early hematopoietic development in vertebrates. CDX2 is not normally expressed in adult murine and human normal hematopoiesis, but is reactivated in 90% of AML patients (Scholl et al., 2007). Aberrant expression of CDX2 in bone marrow stem and progenitor cells results in down-regulation of KLF4 expression as the putative leukemogenic event (Faber et al., 2013). LOR-253 induces KLF4 expression in AML, leading to cell death by apoptosis.

Anticancer activity of LOR-253 in vitro

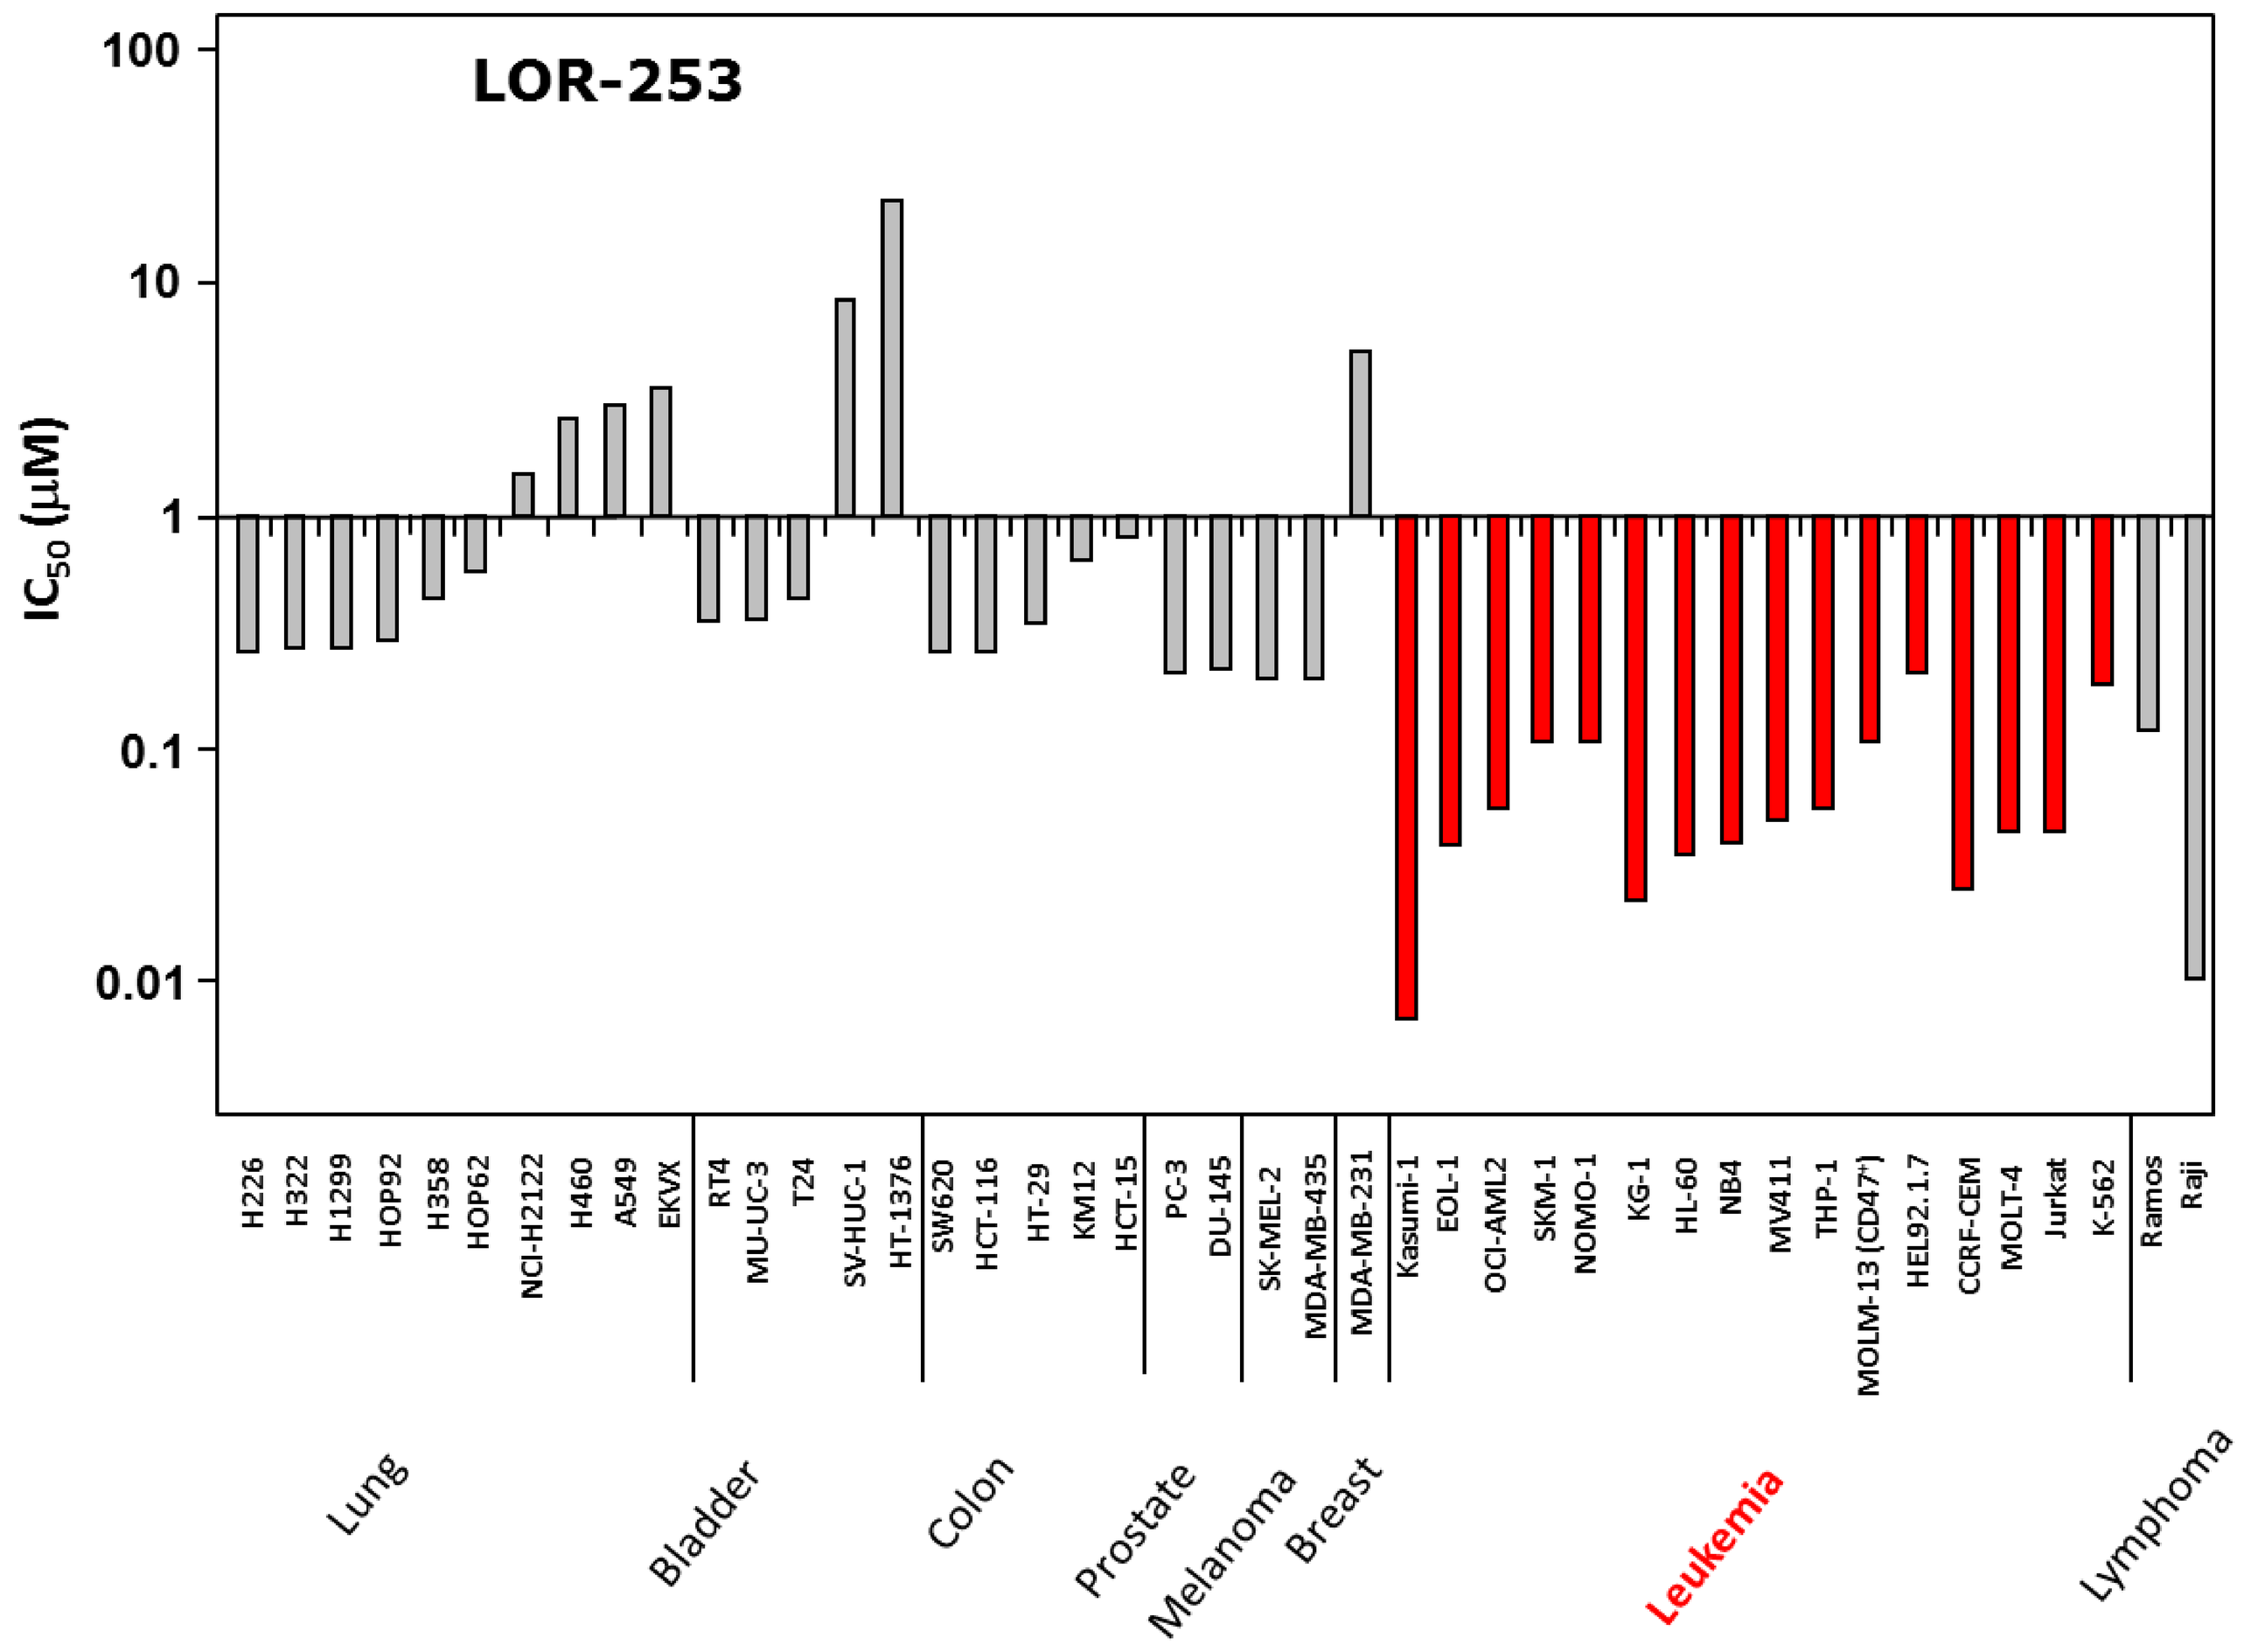
LOR-253
IC_{50} (µM)
100
10
1
0.1
0.01
H226
H322
H1299
HOP92
H358
HOP62
NCI-H2122
H460
A549
EKVX
RT4
MU-UC-3
T24
SV-HUC-1
HT-1376
SW620
HCT-116
HT-29
KM12
HCT-15
PC-3
DU-145
SK-MEL-2
MDA-MB-435
MDA-MB-231
Kasumi-1
EOL-1
OCI-AML2
SKM-1
NOMO-1
KG-1
HL-60
NB4
MV411
THP-1
MOLM-13 (CD47⁺)
HEL92.1.7
CCRF-CEM
MOLT-4
Jurkat
K-562
Ramos
Raji
Lung
Bladder
Colon
Prostate
Melanoma
Breast
Leukemia
Lymphoma

Figure 2. Antiproliferative activity of LOR-253 in solid tumor, leukemia and lymphoma cell lines. Leukemia and lymphoma cells (4 × 10³/well) in 50 µL of growth medium were seeded in 96-well cell culture plates and incubated overnight at 37° C, followed by the addition of 50 µL of medium containing LOR-253 or 0.1% DMSO. For solid tumor cell lines, 2 × 10³ cells/well in 100 µL of growth medium were seeded in 96-well cell culture plates and incubated overnight at 37° C. The medium was removed and replaced with a total volume of 100 µL growth medium containing LOR-253 or 0.1% DMSO. After incubation at 37° C for 5 days, cell viability was quantitated with the use of an XTT colorimetric assay. The plates were further incubated at 37° C for 4 h, and the absorbance of each well was measured at 490 nm. The data from four independent experiments were adjusted relative to the blank and expressed as a percentage of cell growth compared with the DMSO control. Values shown are mean concentrations of LOR-253 that inhibit cell growth by 50% (IC_{50}).

LOR-253 induces KLF4 and p21 in AML

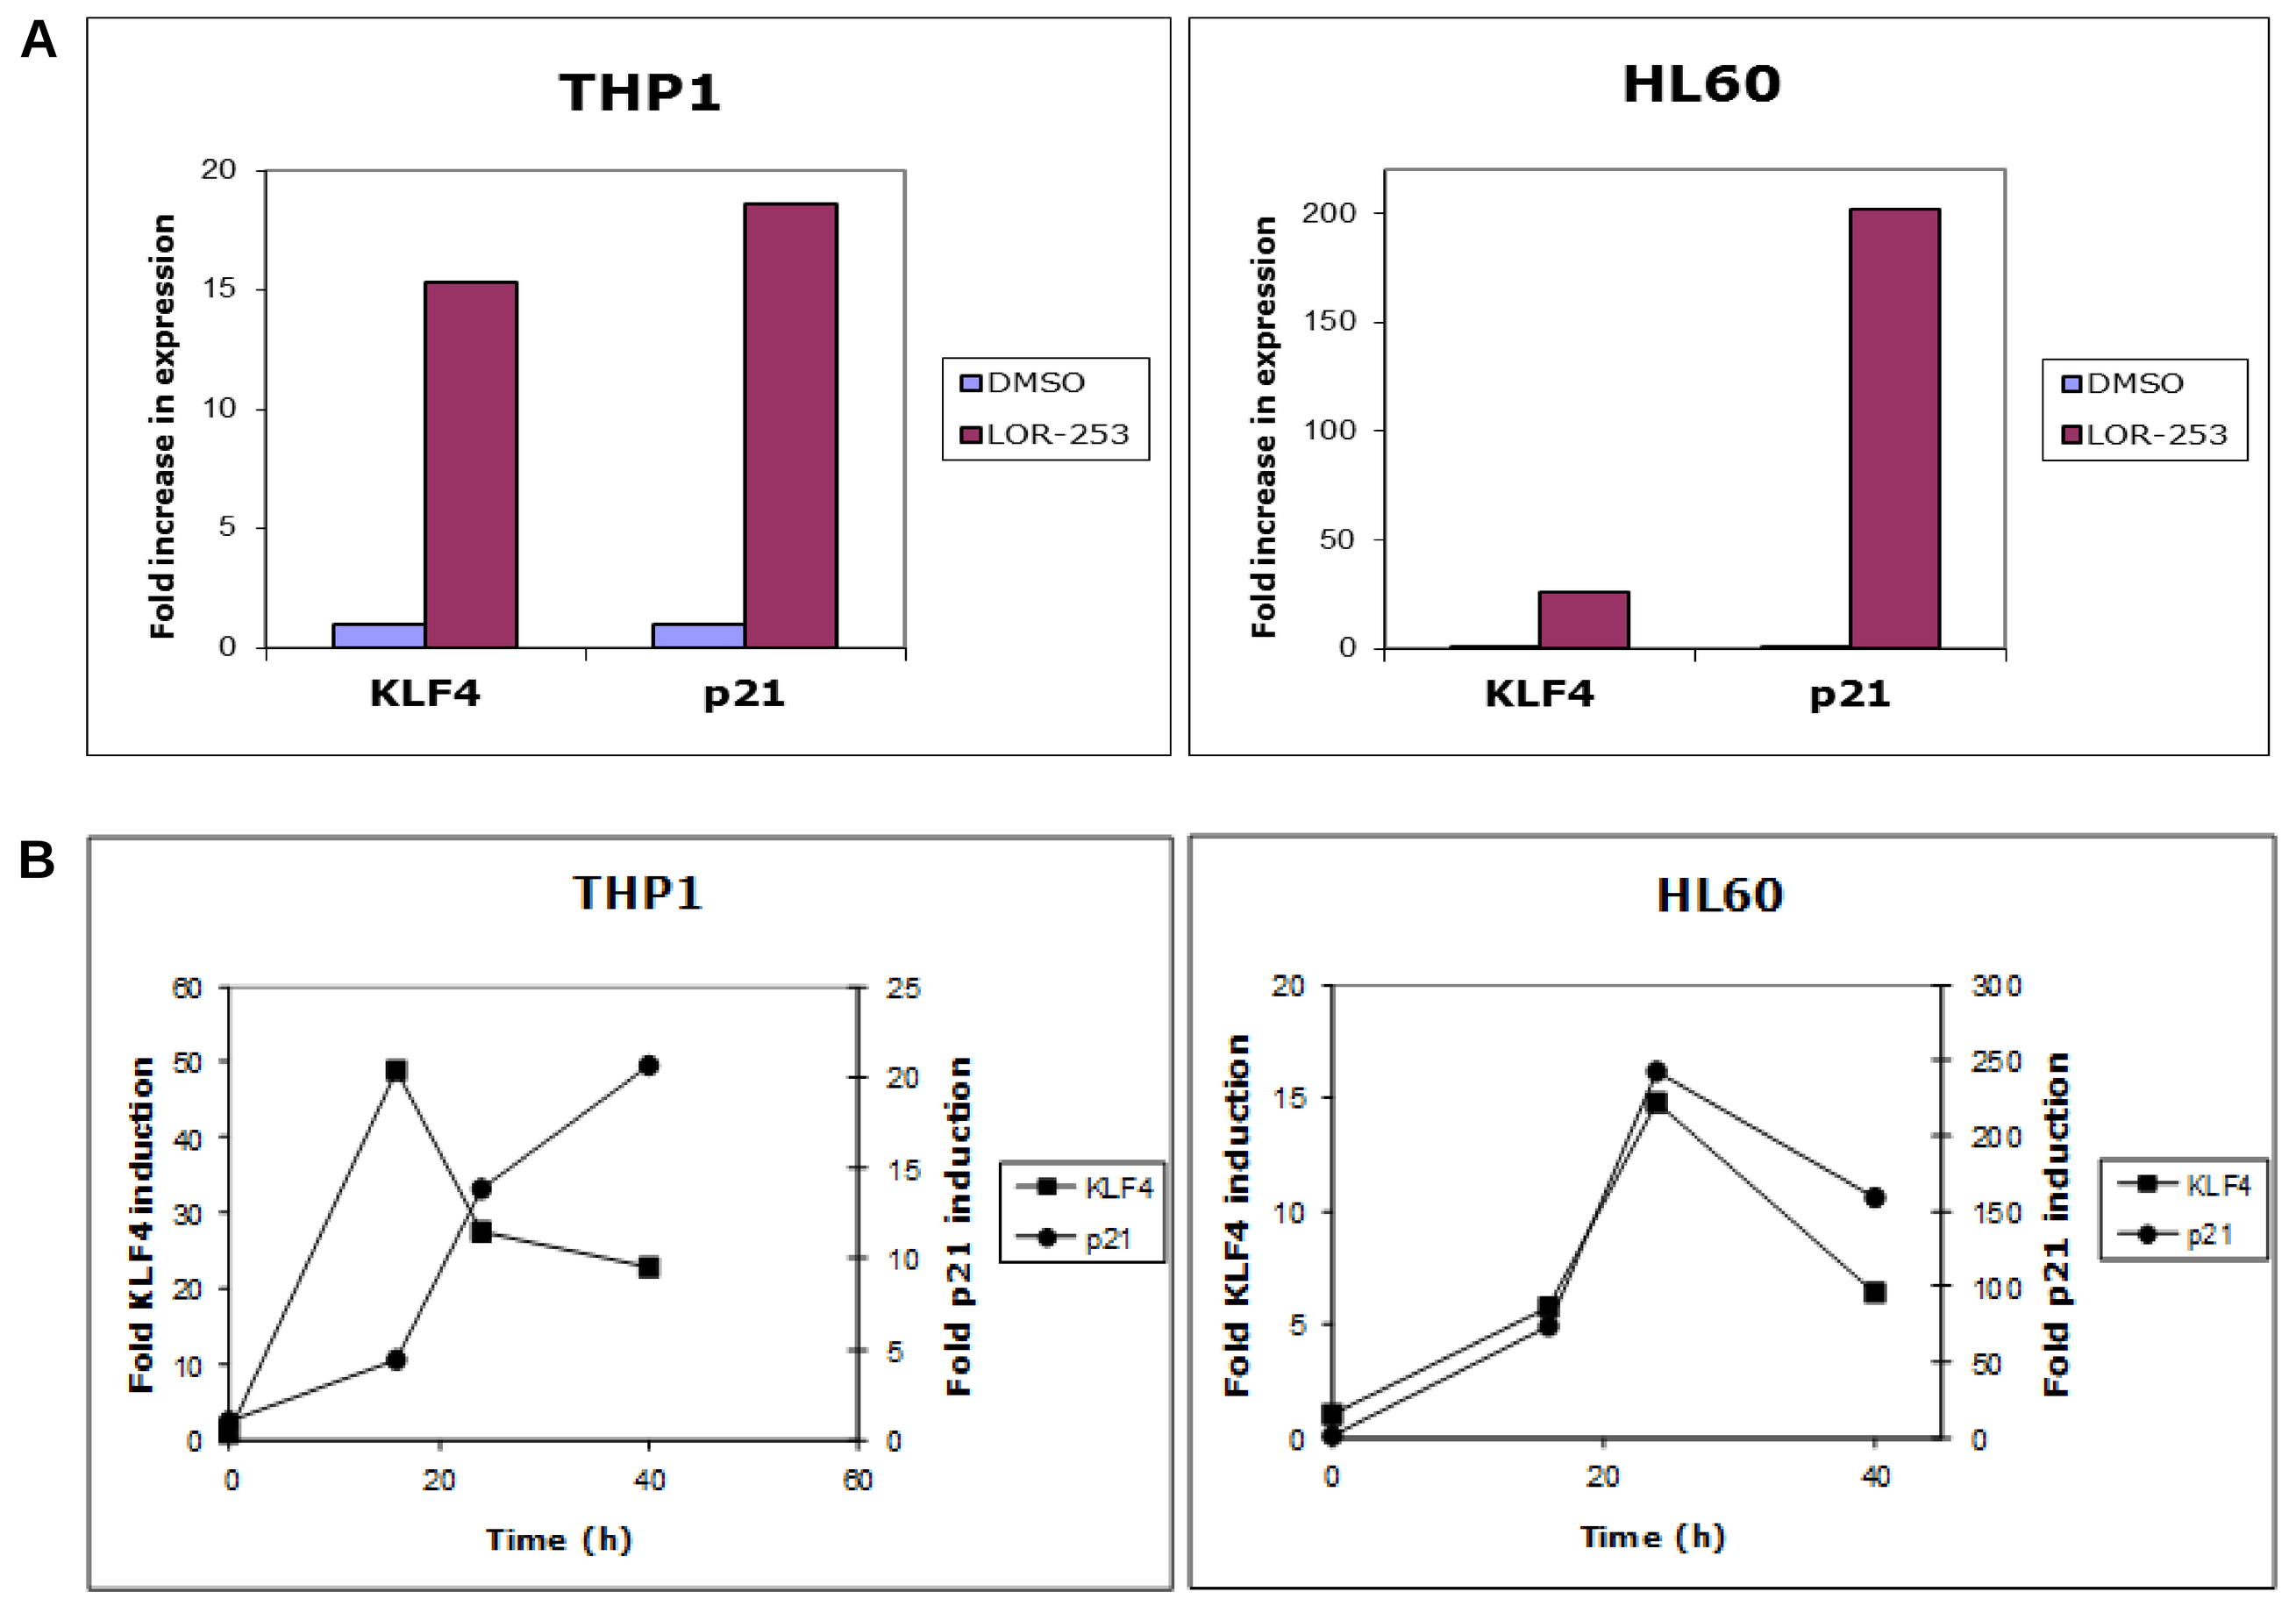
A
THP1
HL60
Fold increase in expression
DMSO
LOR-253
KLF4
p21
B
THP1
HL60
Fold KLF4 induction
Fold p21 induction
Time (h)
KLF4
p21

Figure 3. LOR-253 induces KLF4 and p21 expression in AML. (A) AML cell lines THP1 and HL60 were treated with 0.5 µM LOR-253 or DMSO for 16 h and expression of KLF4 and p21 was determined by quantitative RT-PCR. (B) Kinetics of KLF4 and p21 expression in THP1 and HL60 in response to LOR-253 treatment. Cells were treated with DMSO or 0.5 µM LOR-253 and analyzed for KLF4 and p21 expression over a time course up to 40 h. KLF4 expression peaked at 16 – 24h in both cell lines. By contrast, p21 levels increased steadily in THP1 up to 40 h and tracked closely with KLF4 expression in HL60. For all studies KLF4 and p21 expression levels were normalized to β-actin.

LOR-253 induces G1 arrest and apoptosis in AML

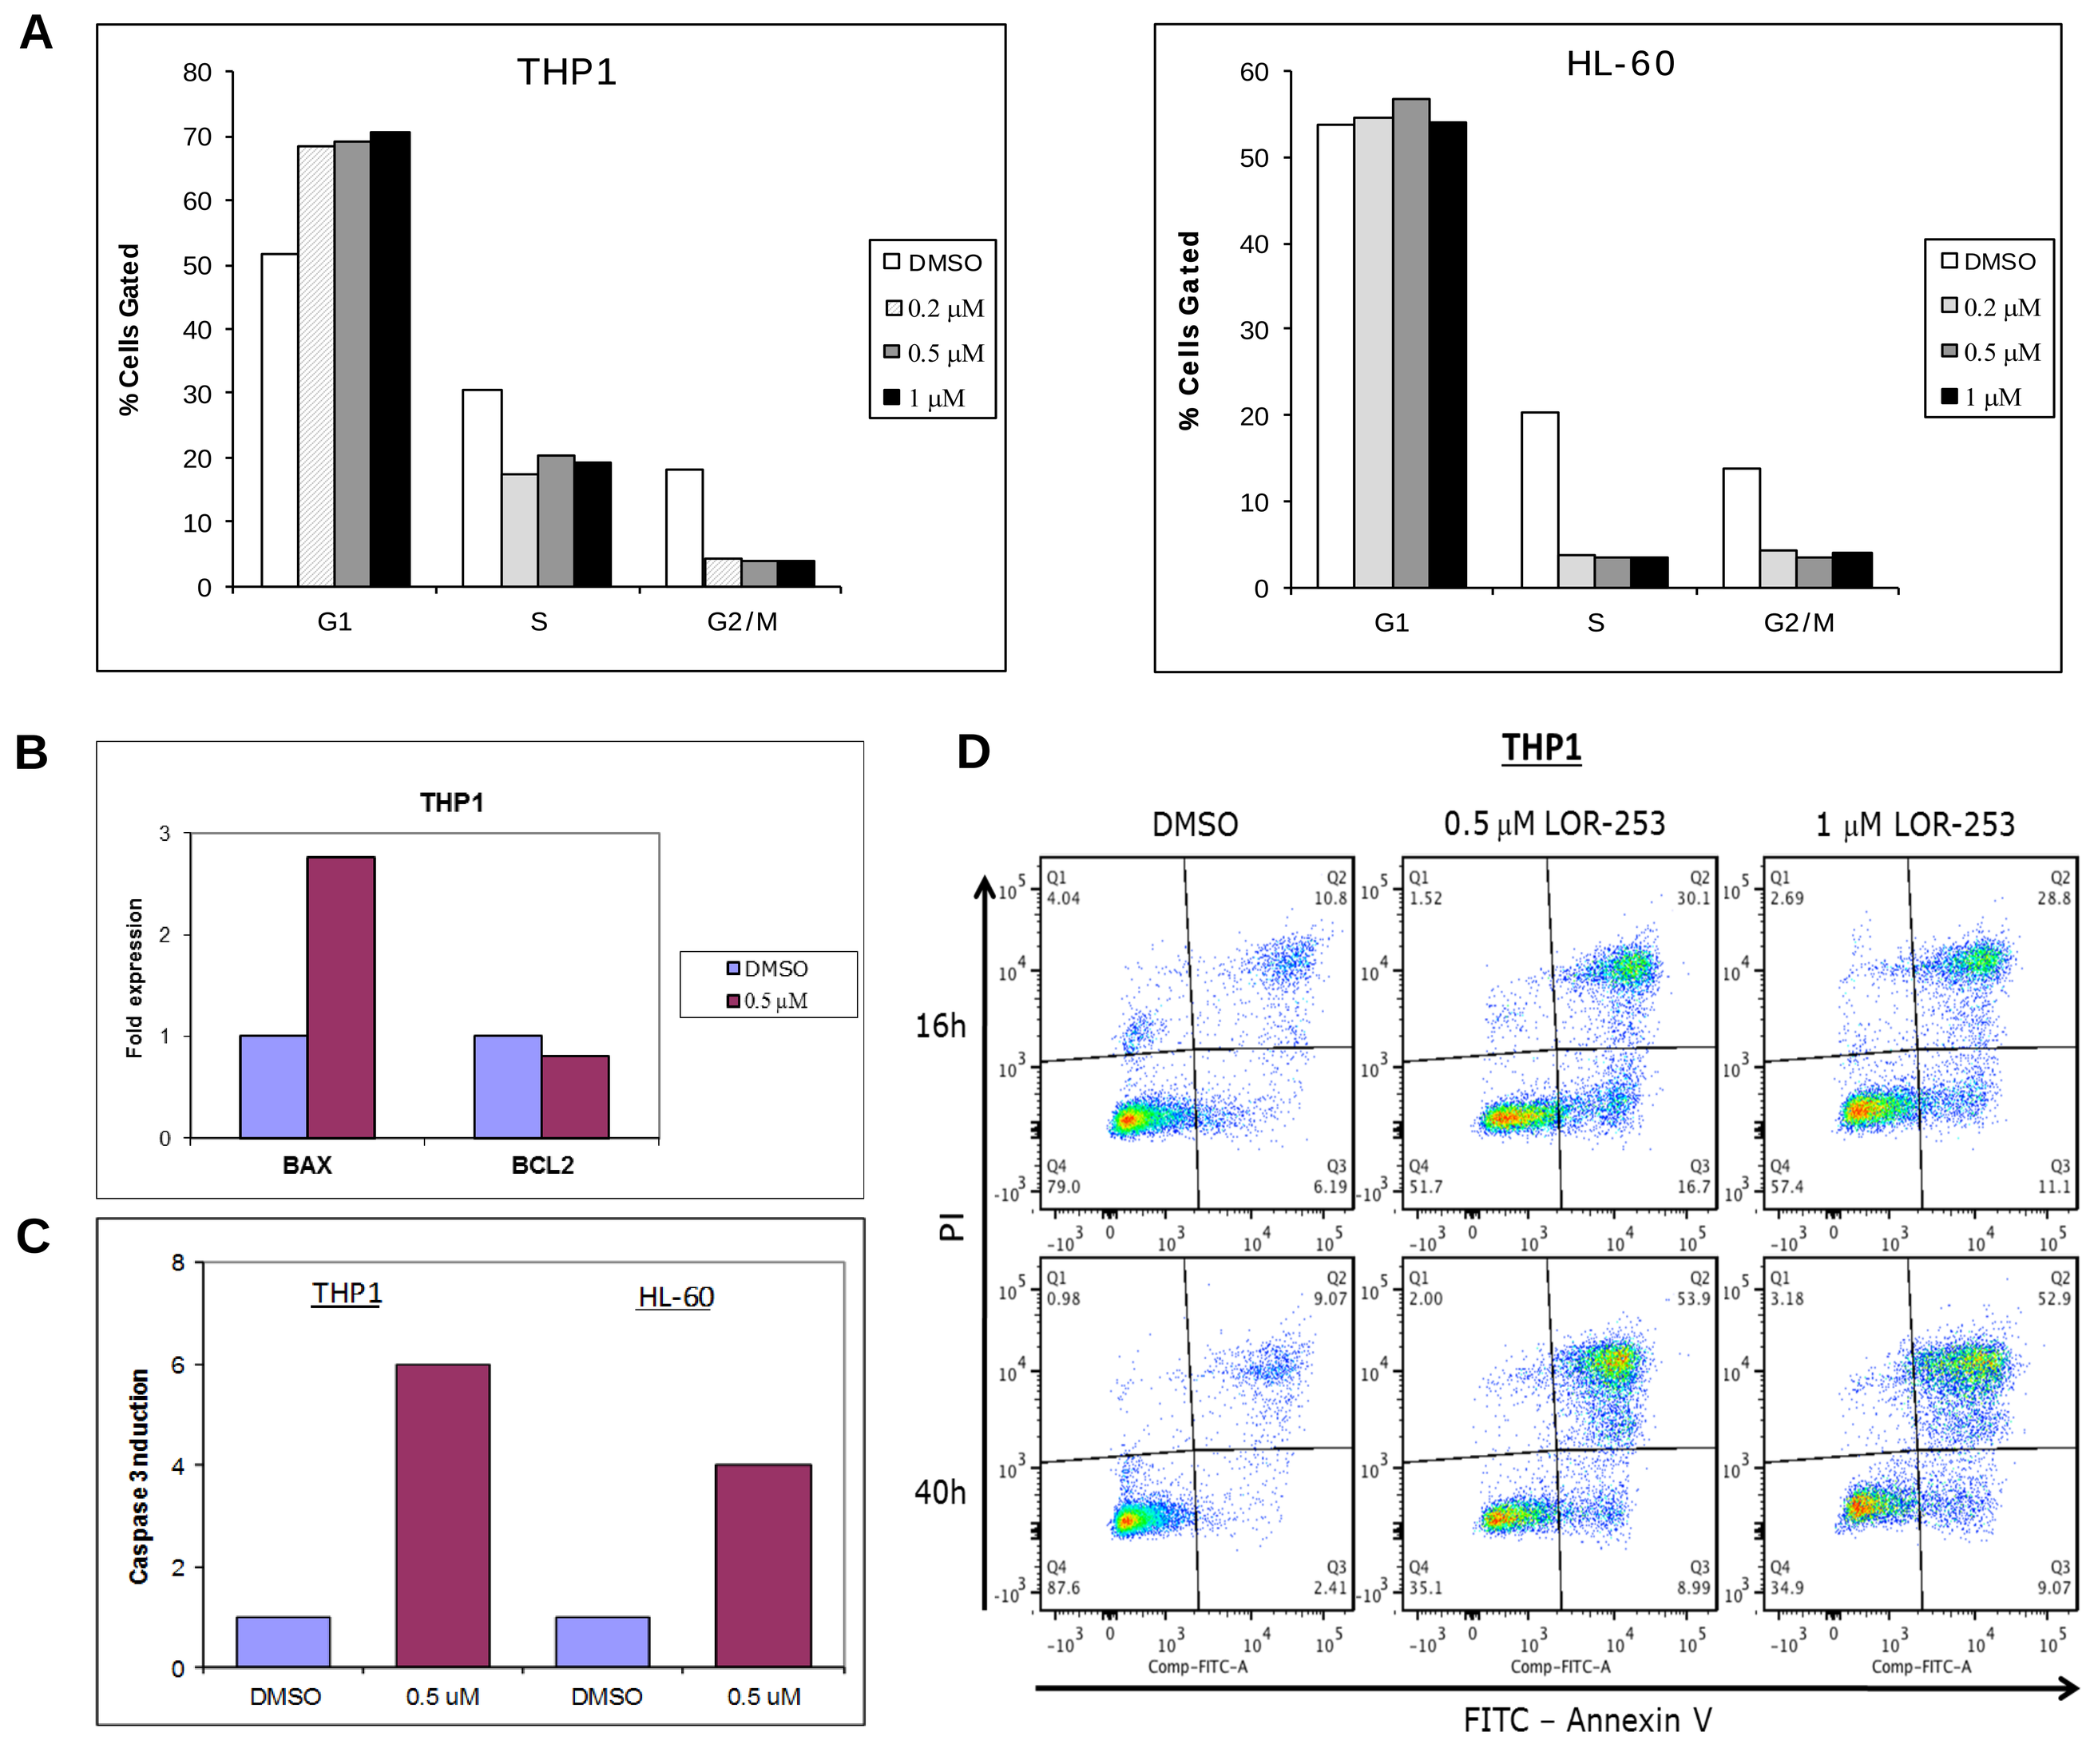
A
THP1
HL-60
% Cells Gated
DMSO
0.2 µM
0.5 µM
1 µM
G1
S
G2/M
B
THP1
Fold expression
BAX
BCL2
DMSO
0.5 µM
C
THP1
HL-60
Caspase 3 Induction
DMSO
0.5 uM
D
THP1
DMSO
0.5 µM LOR-253
1 µM LOR-253
16h
40h
PI
FITC – Annexin V

Figure 4. (A) Effect of LOR-253 on cell cycle in AML. THP1 and HL-60 cells were treated with increasing concentrations of LOR-253 for 16 h and stained with propidium iodide (PI) followed by cell cycle analysis with flow cytometry. Both cell types showed cell cycle arrest at G1 phase with LOR-253 treatment compared to DMSO control. (B) LOR-253 induces apoptosis in AML. THP1 cells treated with 0.5 µM LOR-253 for 48 h showed upregulation of pro-apoptotic gene BAX and decreased expression of anti-apoptotic BCL2 by qRT-PCR. (C) Treatment of THP1 and HL-60 cells with 0.5 µM for 48 h resulted in high level induction of caspase 3 activity compared to DMSO control. (D) THP1 cells were treated with 0.5 – 1 µM LOR-253 or DMSO for 16 – 40 h, followed by double staining with Annexin V-FITC and PI. LOR-253 treated cells showed increased levels of apoptotic cells (Annexin V+/PI-; Annexin V+/PI+)

Anticancer synergy of LOR-253 in AML

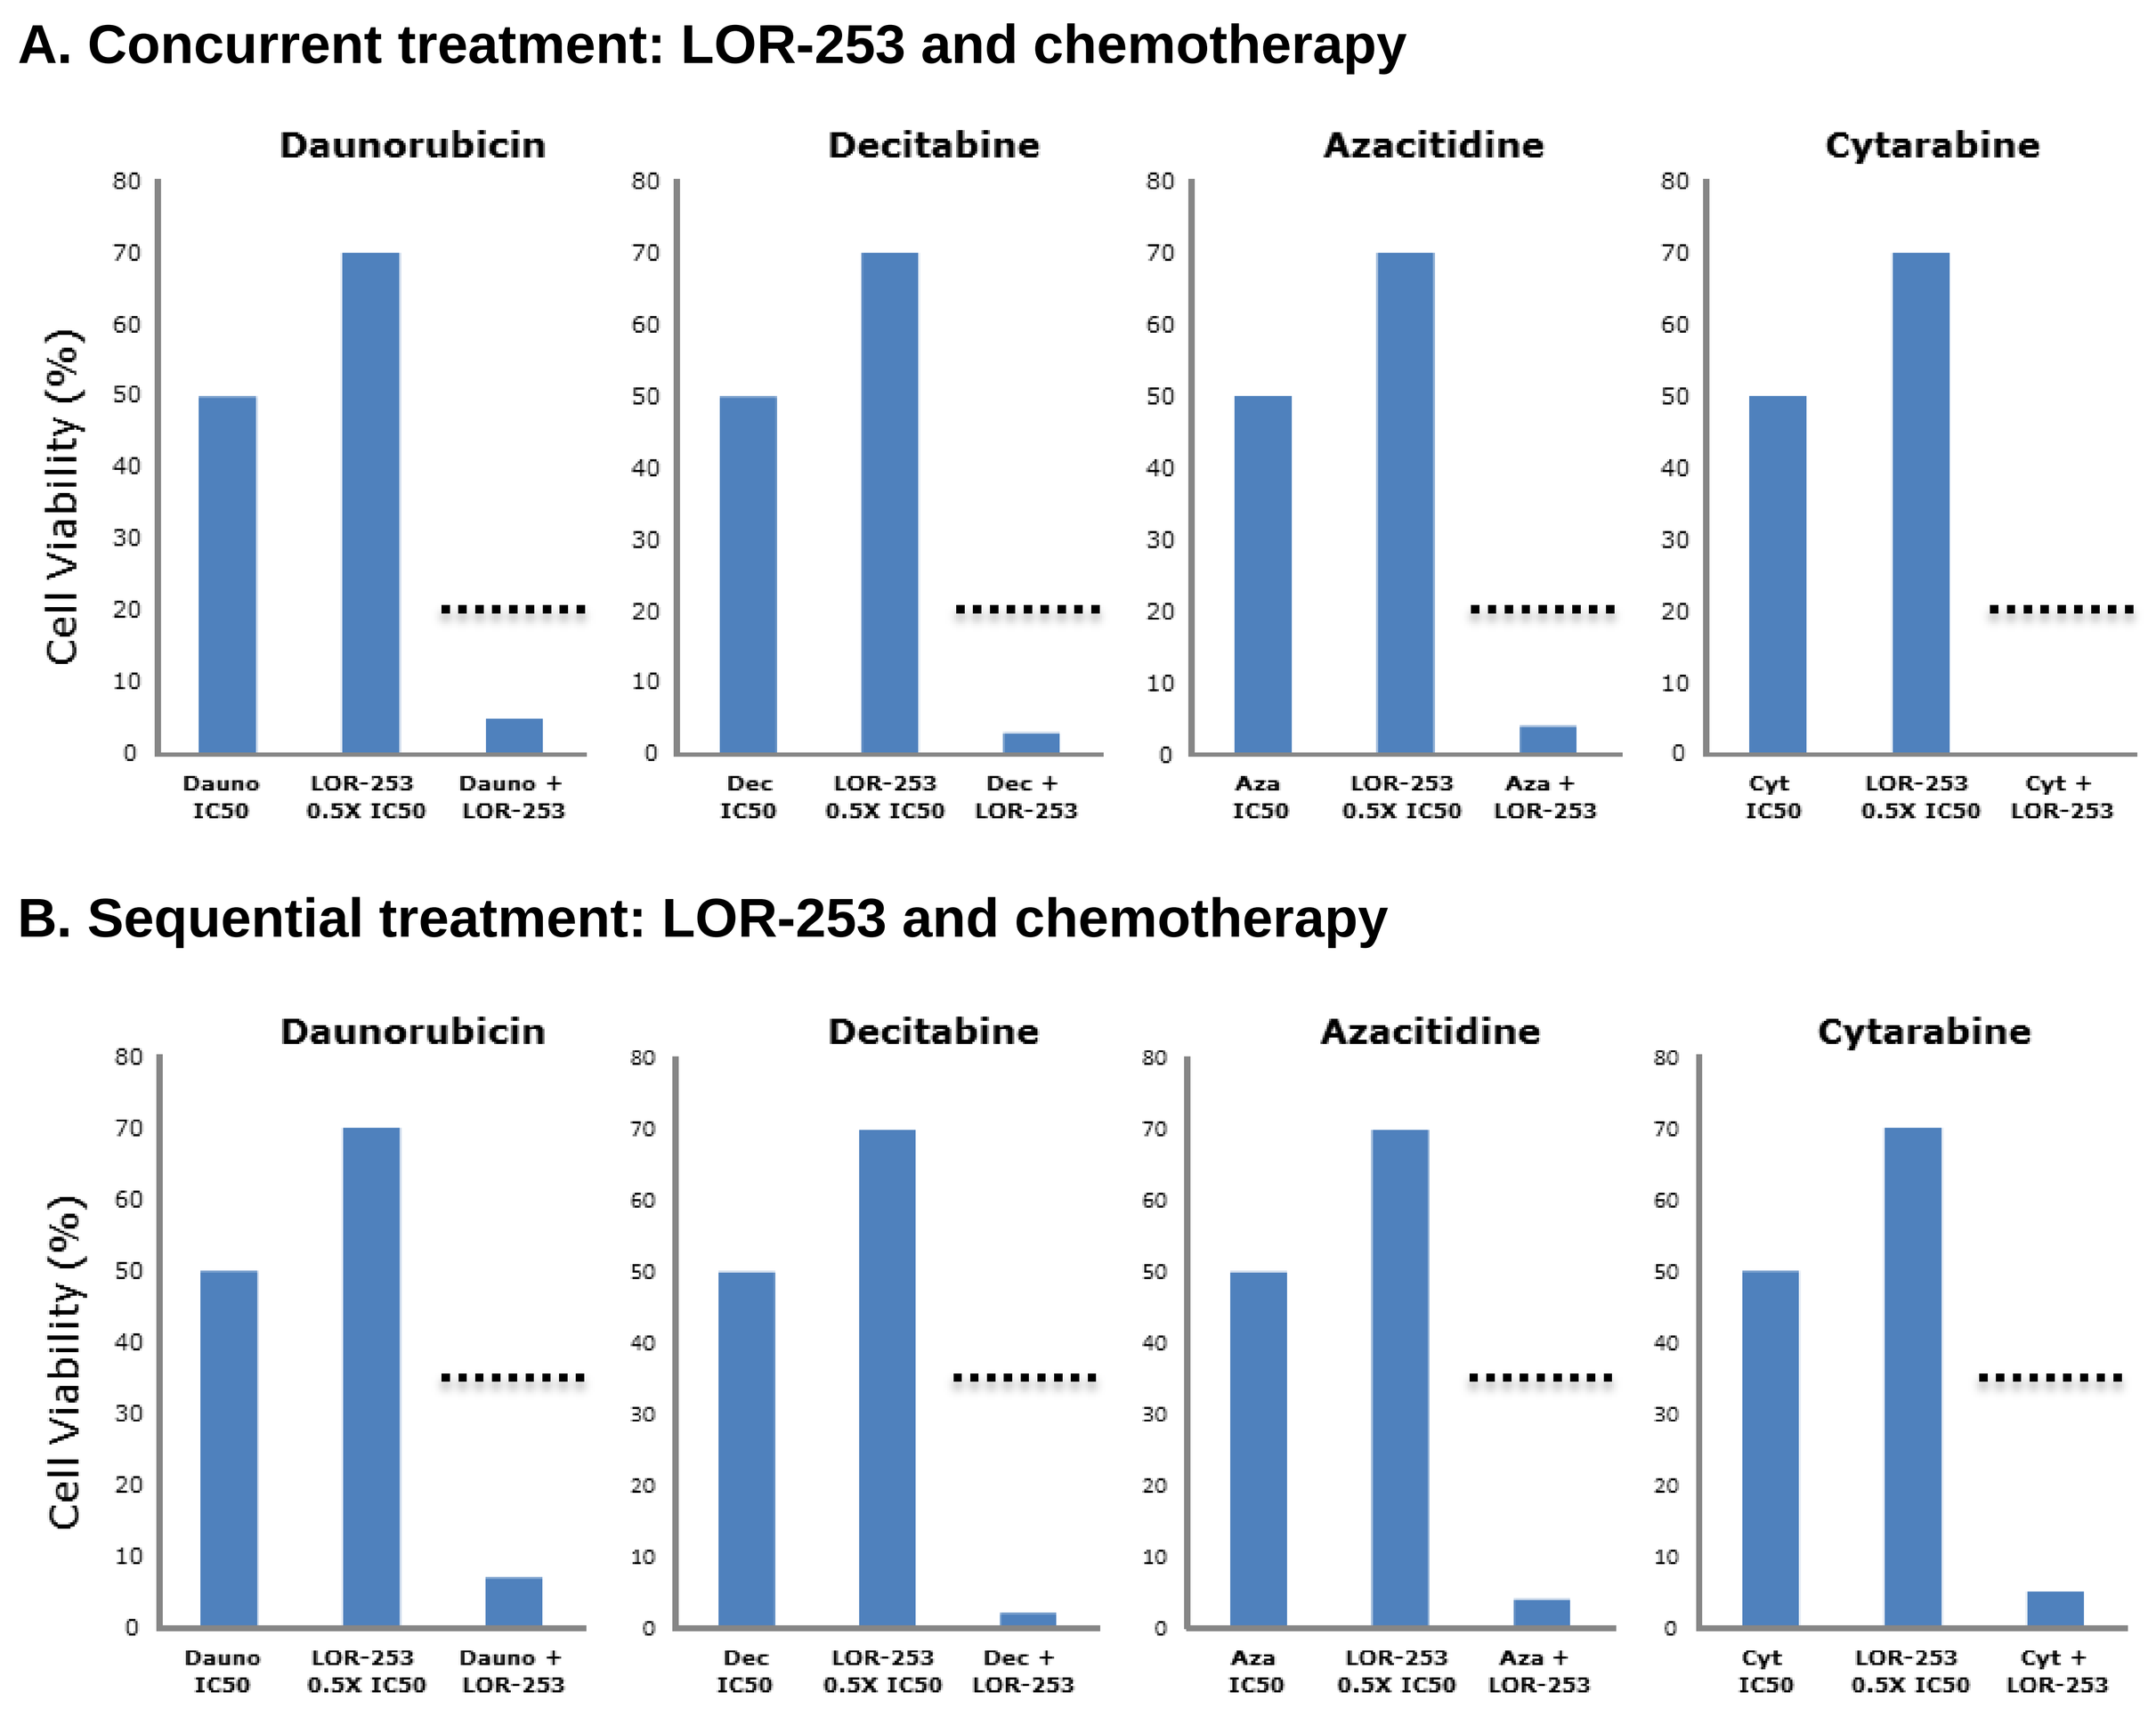
A. Concurrent treatment: LOR-253 and chemotherapy
Daunorubicin
Decitabine
Azacitidine
Cytarabine
Cell Viability (%)
Dauno IC50
LOR-253 0.5X IC50
Dauno + LOR-253
Dec IC50
Dec + LOR-253
Aza IC50
Aza + LOR-253
Cyt IC50
Cyt + LOR-253
B. Sequential treatment: LOR-253 and chemotherapy

Figure 5. LOR-253 synergizes with AML chemotherapy drugs. (A) For concurrent treatment, HL-60 cells (6 × 10³/well) were seeded in 96-well plates and treated with chemotherapy plus LOR-253 for five days. (B) For sequential treatment, HL-60 cells were treated with chemotherapy for two days. Cells were then washed and resuspended in media containing LOR-253 and incubated for three days. Chemotherapies were tested at IC_{50} concentrations (daunorubicin, 0.0058 µM; decitabine, 0.12 µM; azacitidine, 3.53 µM; cytarabine, 0.22 µM), while LOR-253 was tested at 0.5X IC50 (0.02 µM). Cell viability was determined using an XTT assay as described in Fig. 2. The dashed line (----) indicates the predicted viability if the combination effect was additive.

Summary

- Approximately **90% of patients with AML aberrantly express CDX2** gene in bone marrow stem and progenitor cells, resulting in **down-regulation of KLF4** expression and contributing to **development of leukemia**
- **LOR-253** is a novel anticancer small molecule and the only **clinical-stage** therapy that **specifically induces expression of the tumor suppressor KLF4** in cancer
- LOR-253 showed **potent antiproliferative activity in a panel of AML** and other leukemia cell lines, with IC_{50} values ranging from 0.007 - 0.3 µM
- Treatment of AML cells with LOR-253 resulted in high level **induction of KLF4 and p21** in a time-dependent manner
- Anticancer activity of LOR-253 in AML is mediated through **cell cycle arrest in G1 and apoptosis**
- LOR-253 has **strong anticancer synergy when used in combination** with AML chemotherapies daunorubicin, azacitidine, decitabine or cytarabine
- Collectively, these preclinical data strongly support further **development of LOR-253 as an AML therapy**

References:

1. Scholl C et al., J Clin Invest. 2007. 117(4):1037-48.
2. Rawat VP et al., Blood. 2008. 111(1):309-19.
3. Faber K et al., J Clin Invest. 2013. 123(1):299-314.