P1-09-08

BCL2L1 (BCLXL) expression and MYC super-enhancer positivity predict sensitivity to the covalent CDK7 inhibitor SY-1365 in Triple Negative Breast Cancer (TNBC) cell lines

Nisha Rajagopal, Graeme Hodgson, Liv Johannessen, Shanhu Hu, Nan Ke, Michael McKeown, Matthew Guenther, Alex Bush, Christian Fritz, David Orlando, Eric Olson, Emmanuelle di Tomaso

Syros Pharmaceuticals, 620 Memorial Drive, Cambridge, MA

Introduction

- CDK7 is an important regulator of transcription (through regulation of RNA Pol II and CDK9) and cell cycle progression (through regulation of cell-cycle kinases CDK1, CDK2, CDK4 and CDK6)
- CDK7 has been implicated in the pathogenesis of multiple malignancies, including cancers of high unmet need such as triple negative breast cancer (TNBC)
- SY-1365, a covalent and selective inhibitor of CDK7, was developed to exploit tumor dependencies driven by CDK7 and is in clinical development in patients with advanced solid tumors.
- Here we report on the in vitro activity of SY-1365 across a large panel of tumor cell lines, including TNBC lines, and identify alterations in mitochondrial apoptosis signaling and epigenetic alterations in the transcriptional regulatory region of *MYC* as potential biomarkers of sensitivity to SY-1365
- In addition, we show that SY-1365 mechanistically down-regulates the mitochondrial apoptosis antagonist MCL1 in TNBC cell lines in vitro and in HCC70 TNBC xenografts in vivo

Schematic of biomarker discovery pipeline

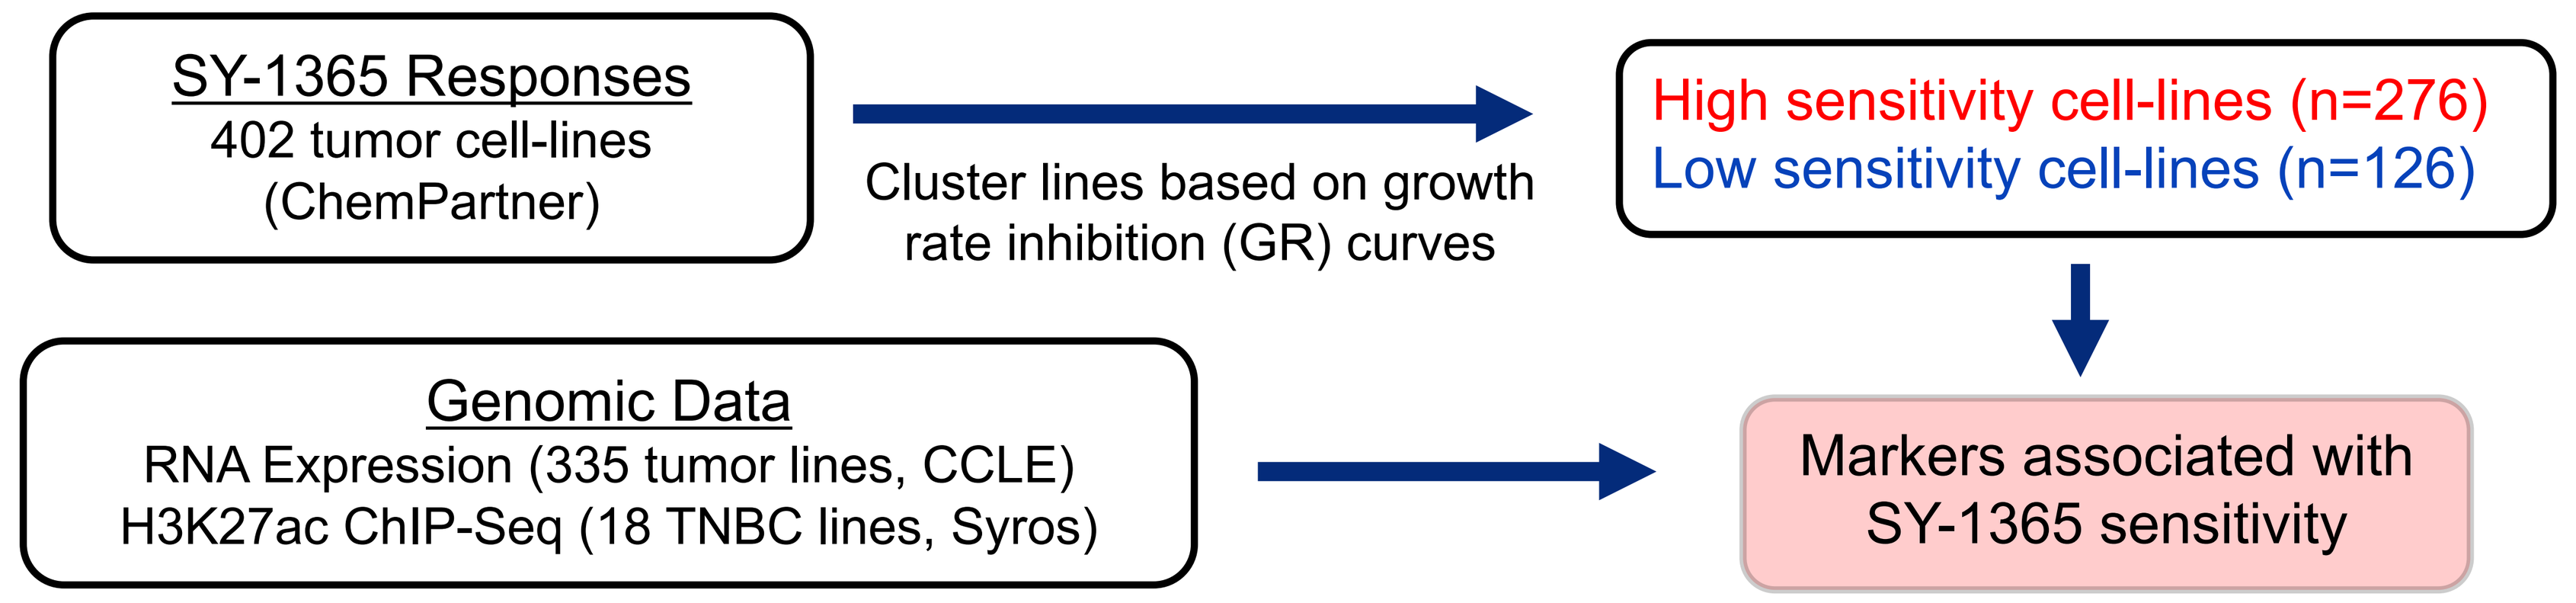

SY-1365 Responses
402 tumor cell-lines
(ChemPartner)
Cluster lines based on growth rate inhibition (GR) curves
High sensitivity cell-lines (n=276)
Low sensitivity cell-lines (n=126)
Genomic Data
RNA Expression (335 tumor lines, CCLE)
H3K27ac ChIP-Seq (18 TNBC lines, Syros)
Markers associated with SY-1365 sensitivity

SY-1365 induces cytotoxic responses in a panel of human tumor cell lines, including in breast cancer cell lines

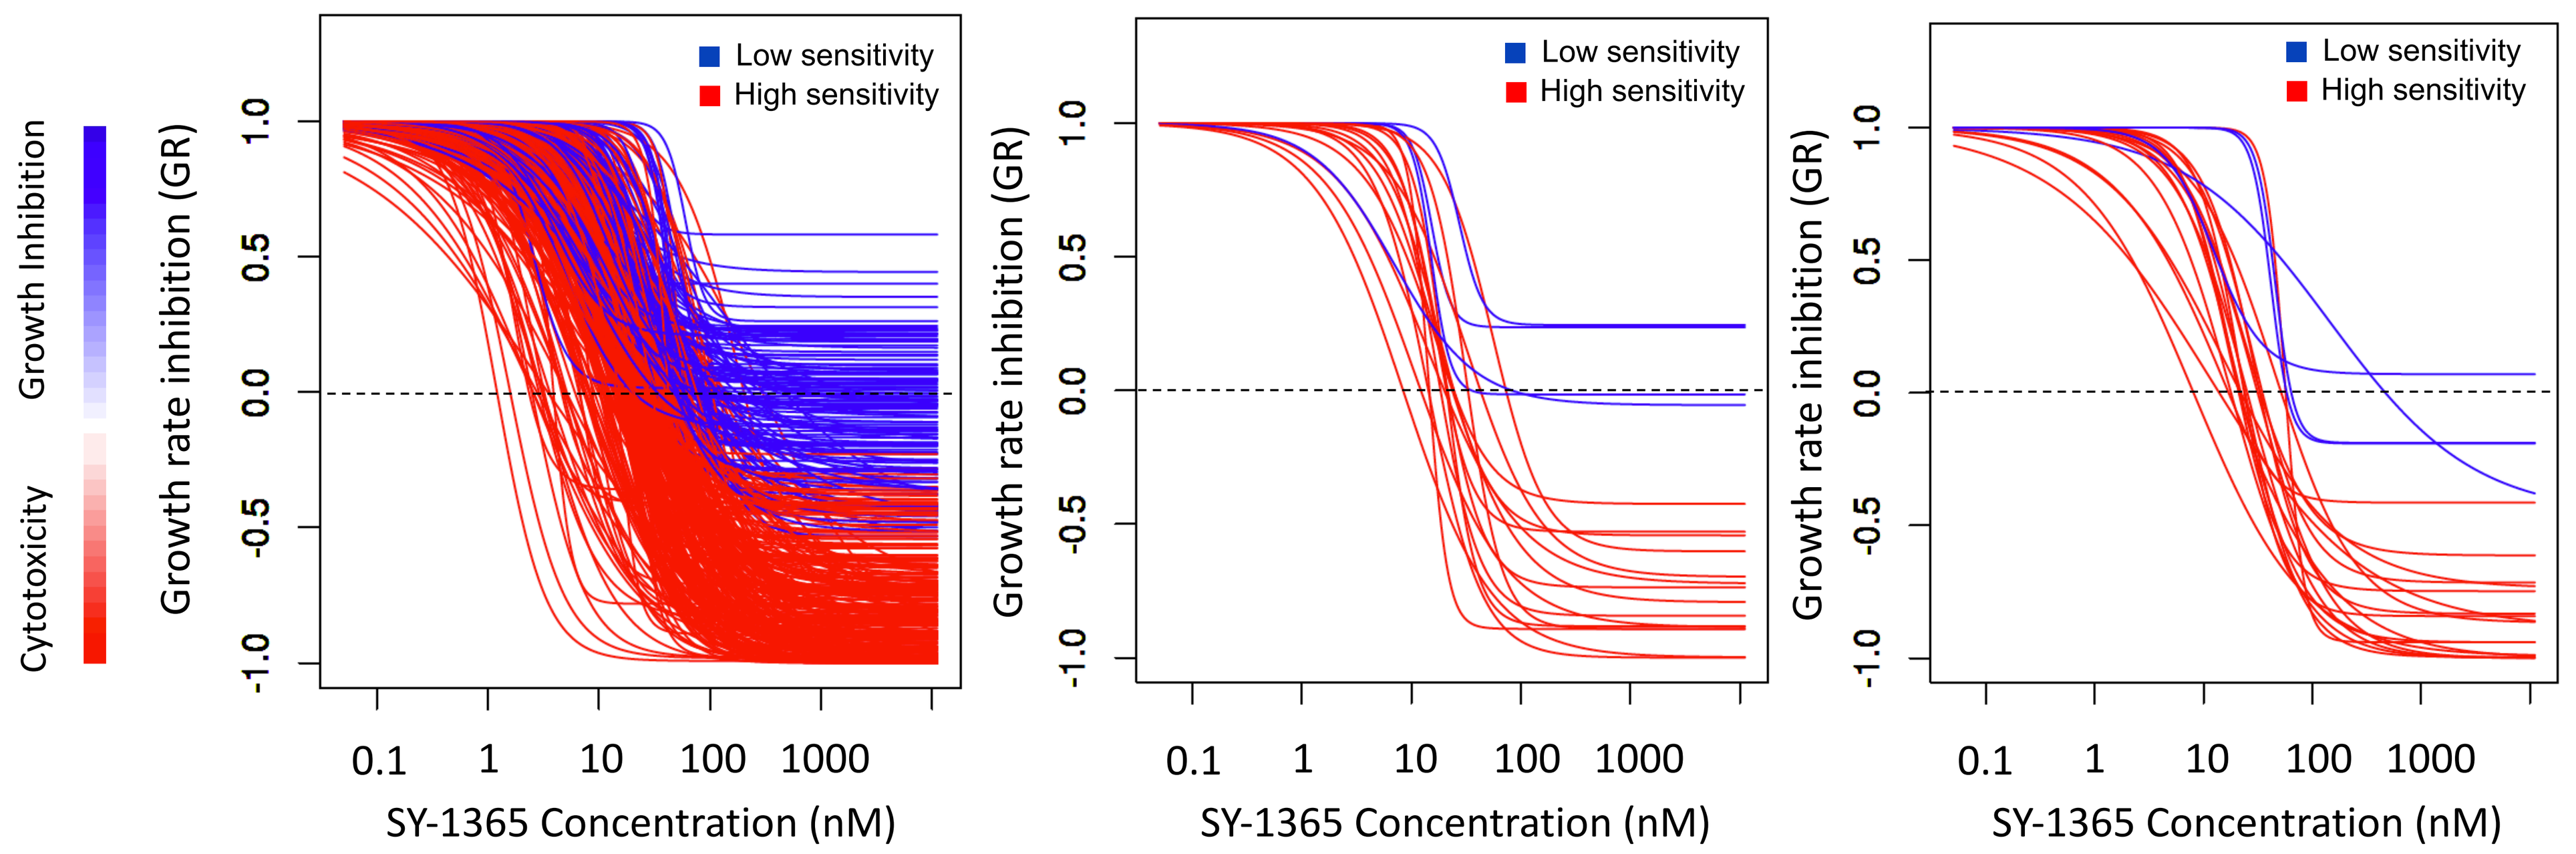

All tumor cell lines (n=402)
TNBC (n=19)
ER+/PR+/HER2+ (n=21)
Low sensitivity
High sensitivity
Growth Inhibition
Cytotoxicity
Growth rate inhibition (GR)
SY-1365 Concentration (nM)

Growth rate inhibition (GR) curves were generated by comparing cell number at baseline to cell number 72h after treatment with SY-1365 at various concentrations (10-point serial dilution). GR<0 indicates cytotoxicity (cell death exceeds cell proliferation). GRmax (depth of response) and GR50 (concentration of SY-1365 required to induce 50% of the maximal effect) were used to classify cell lines into high sensitivity lines (low GRmax, low GR50; red) and low sensitivity lines (high GRmax, high GR50; blue).

SY-1365 sensitivity is associated with low *BCL2L1*/BCLXL expression in TNBC cell lines and PDX models

Low *BCL2L1* expression is the most predictive marker of SY-1365 sensitivity across all cell lines (Accuracy = 70%; FDR=0.006)

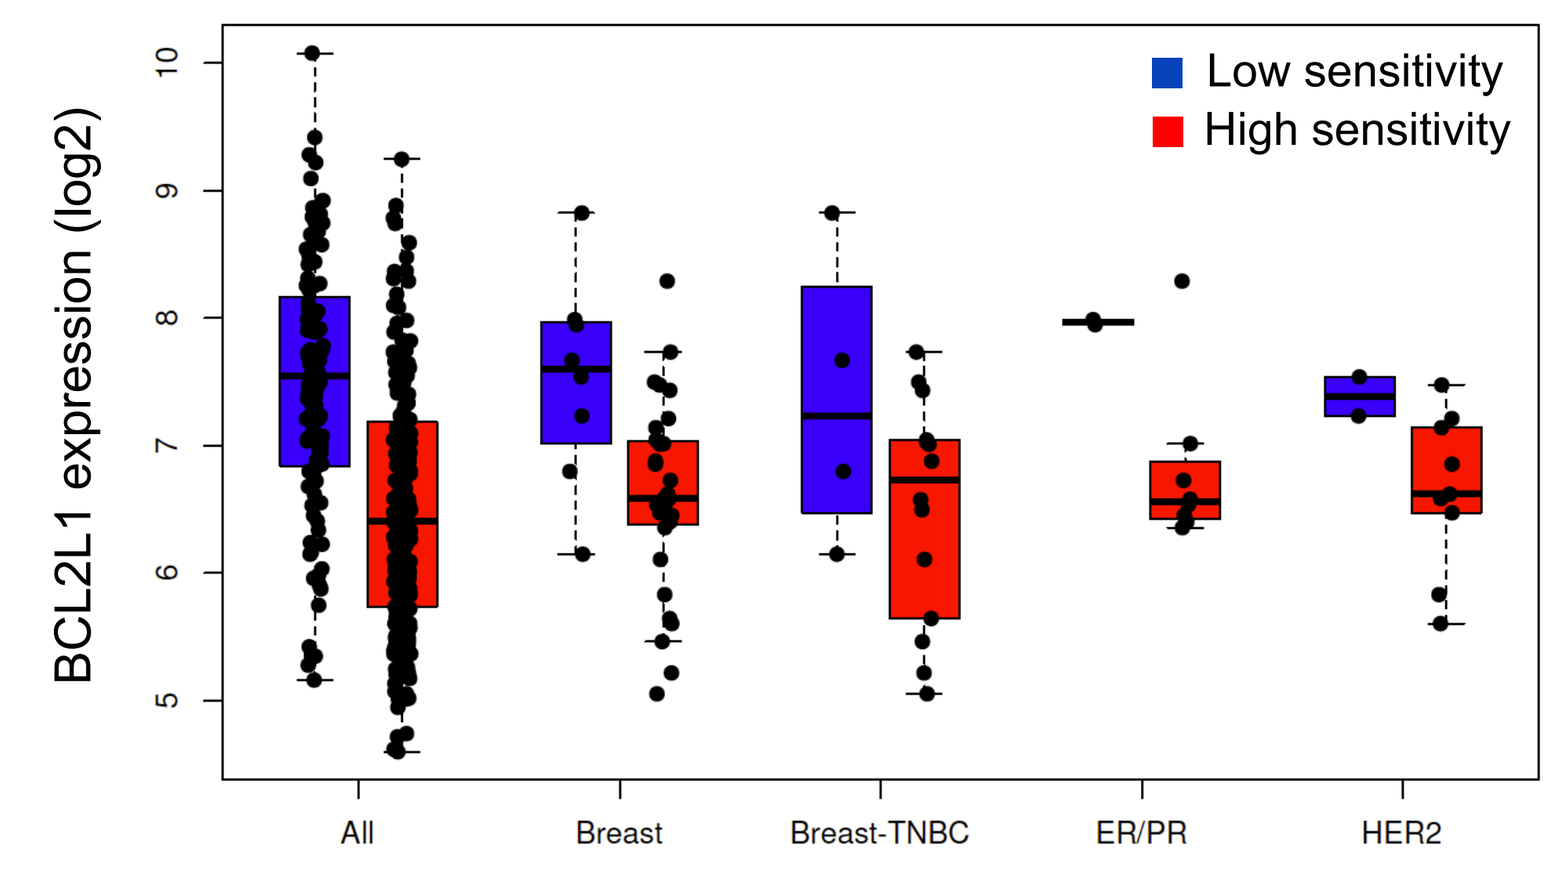

BCL2L1 expression (log2)
Low sensitivity
High sensitivity
All
Breast
Breast-TNBC
ER/PR
HER2

Low baseline BCLXL protein expression is associated with SY-1365 sensitivity in TNBC cell lines

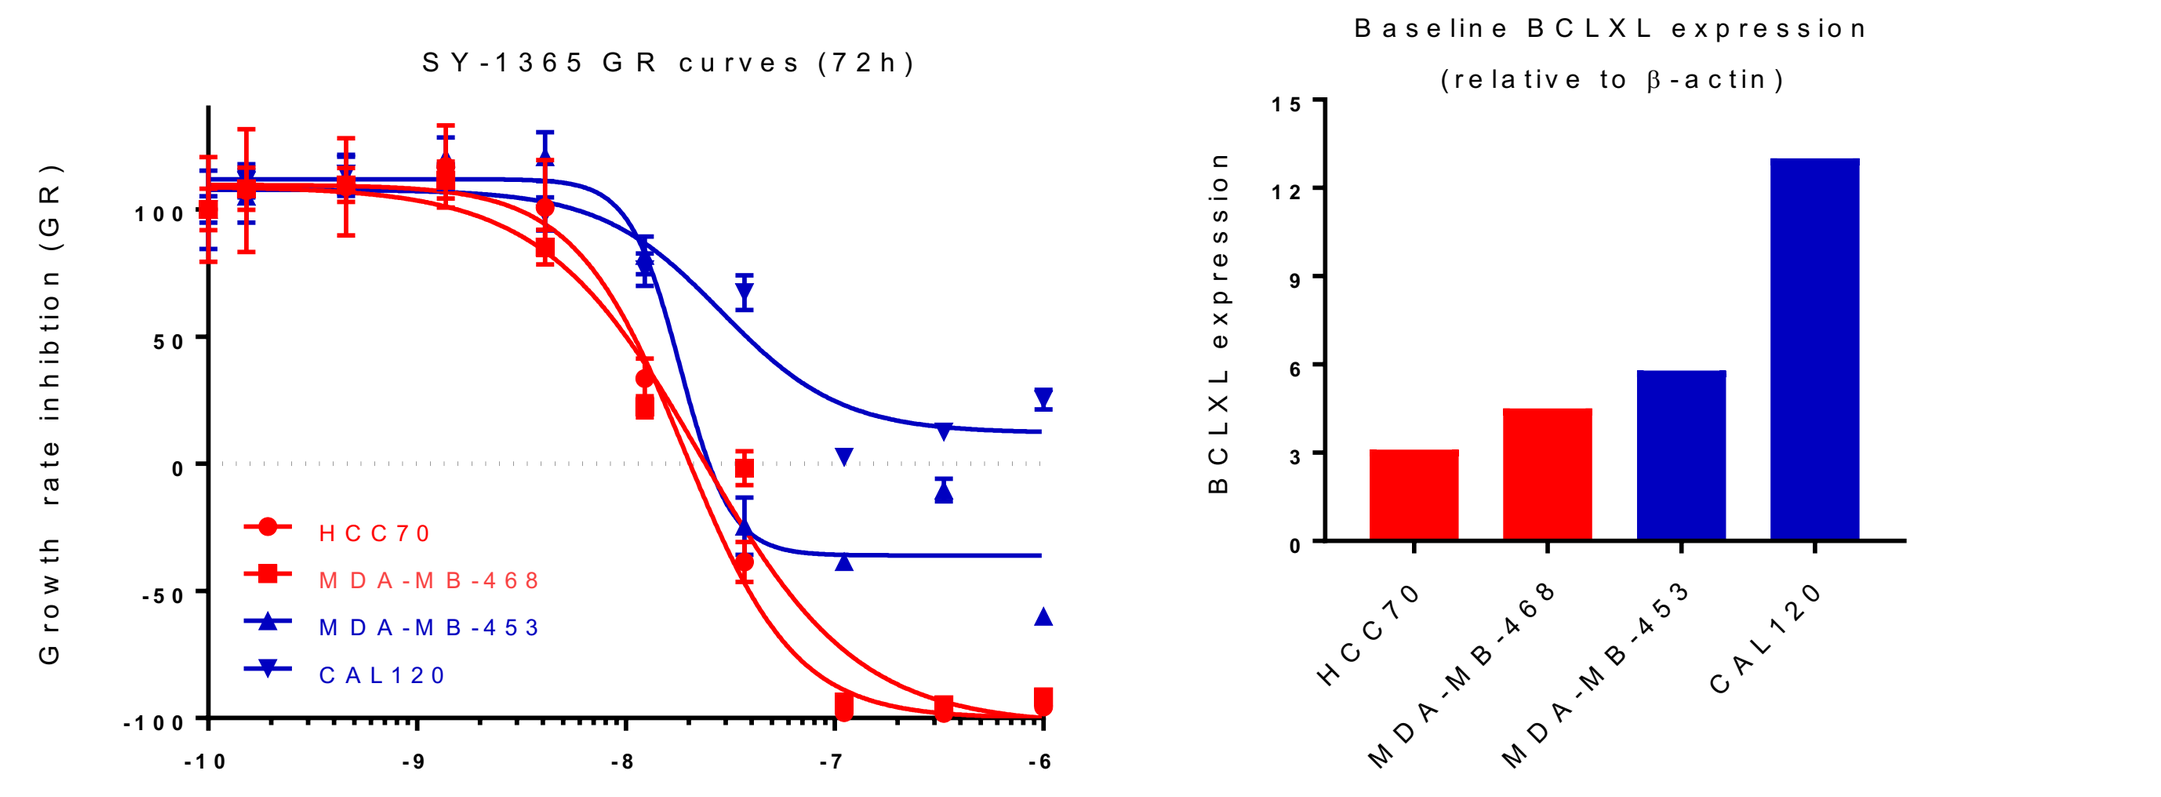

SY-1365 GR curves (72h)
Growth rate inhibition (GR)
HCC70
MDA-MB-468
MDA-MB-453
CAL120
log[M]
Baseline BCLXL expression
(relative to β-actin)
BCLXL expression
HCC70
MDA-MB-468
MDA-MB-453
CAL120

Data derived from densitometry analysis of Western blot shown below

Durable SY-1365 response in TNBC PDXs is associated with low baseline *BCL2L1* expression

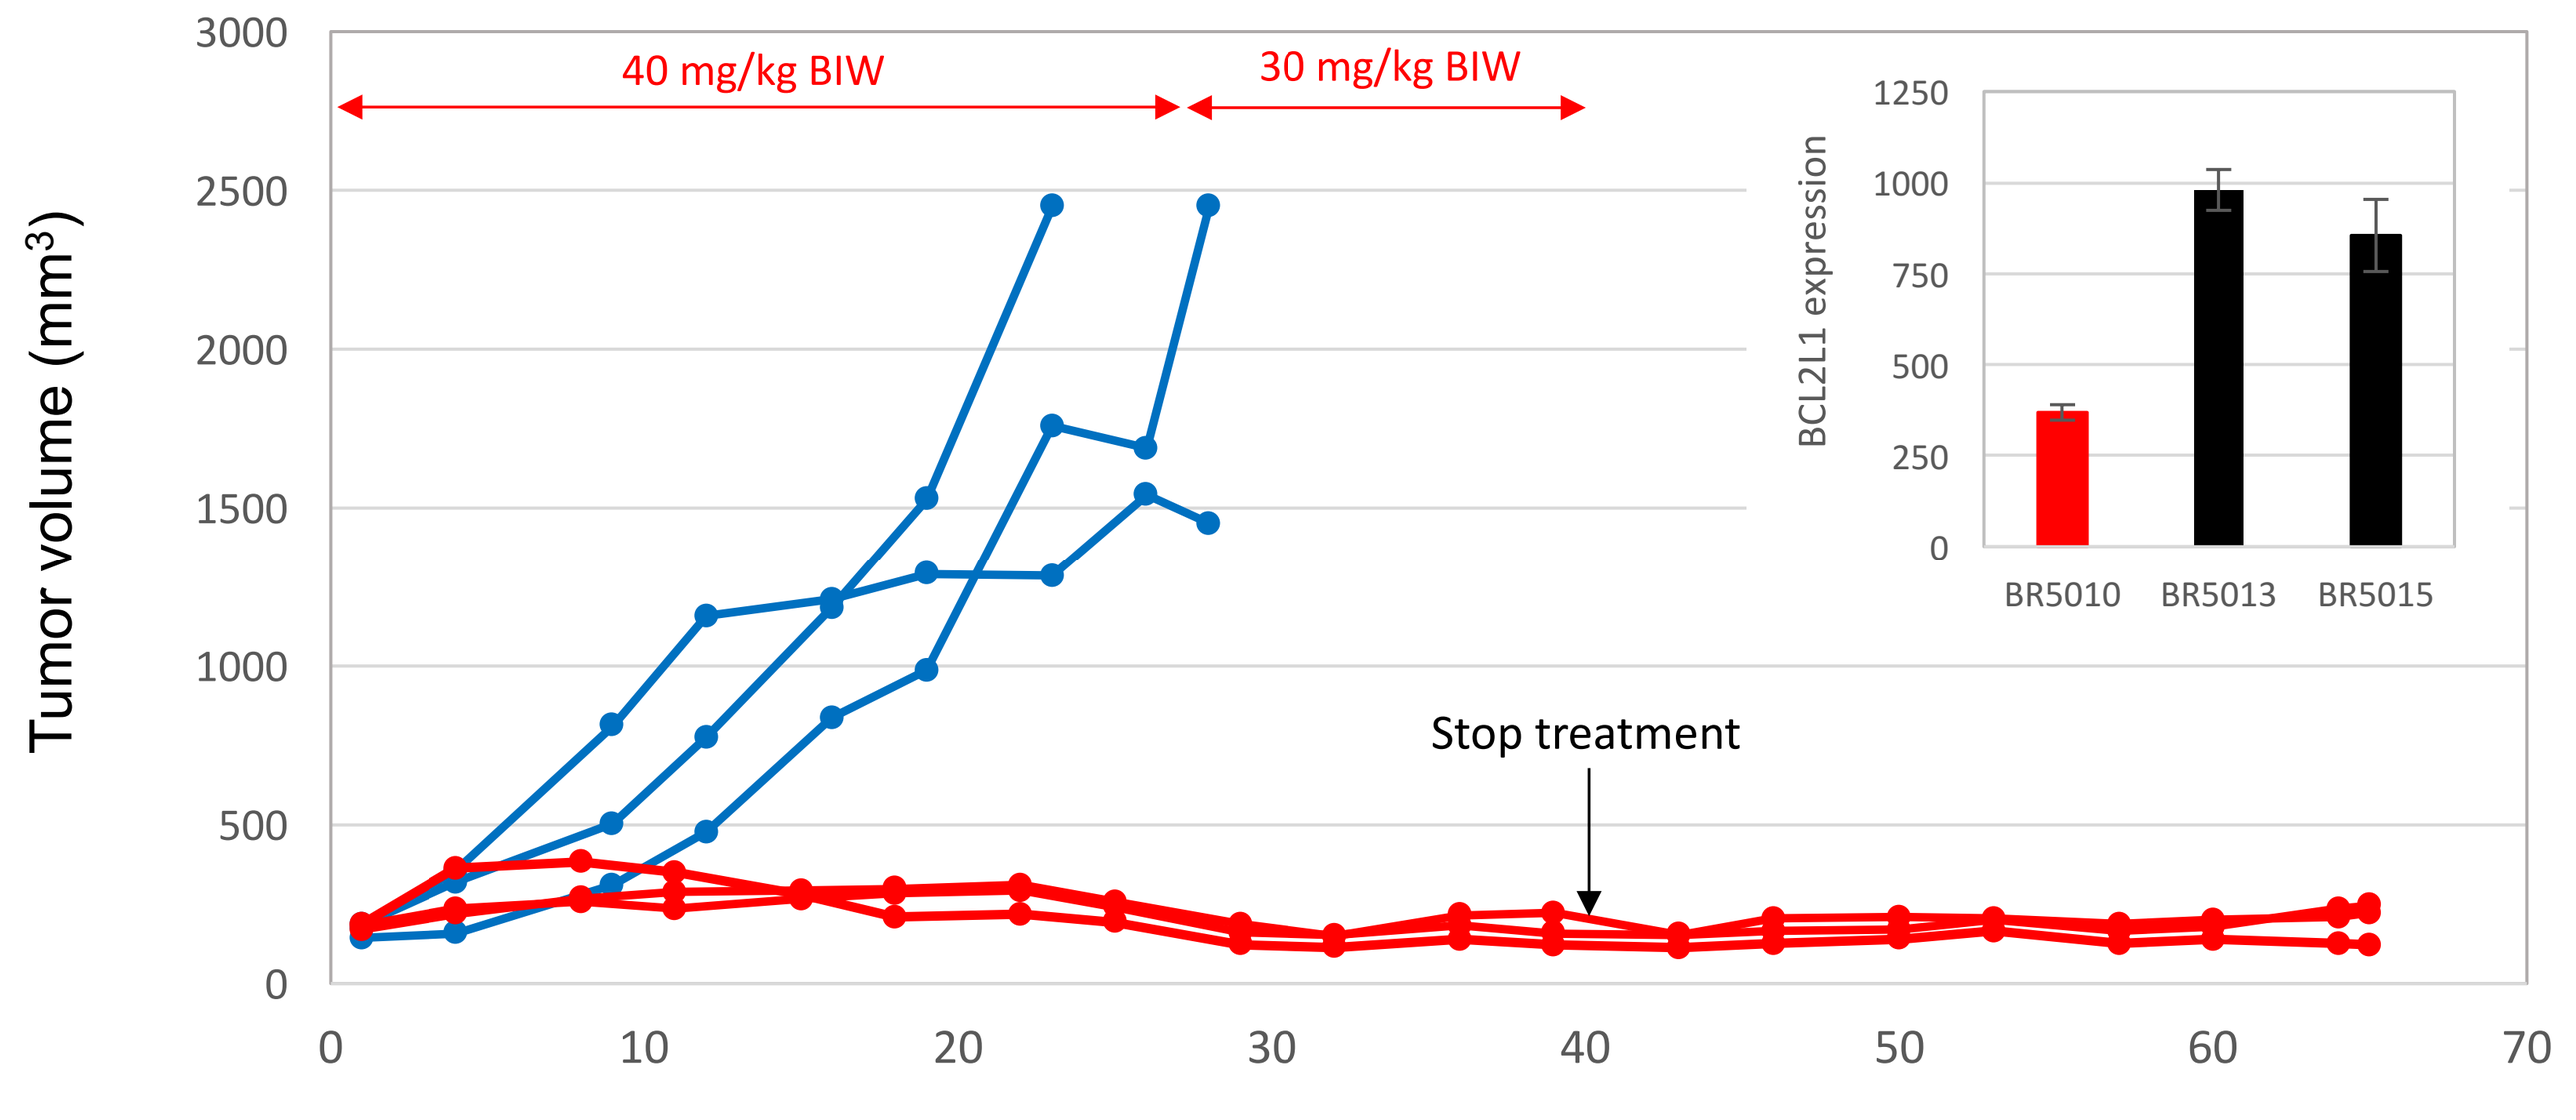

40 mg/kg BIW
30 mg/kg BIW
Tumor volume (mm³)
Stop treatment
Days after treatment start
BCL2L1 expression
BR5010
BR5013
BR5015

Growth of TNBC PDX model BR5010 (Crown Bioscience) during and after treatment with twice weekly (BIW) dosing of SY-1365. Red curves represent tumor growth in individually treated mice (n=3). Blue curves represent historical tumor growth of untreated mice in this model. Inset: Baseline BCL2L1 expression (as measured by Nanostring) in responsive TNBC PDX model BR5010, and non-responsive TNBC PDX models BR5013 and BR5015 (no TGI observed).

Mitochondrial Apoptosis Pathway

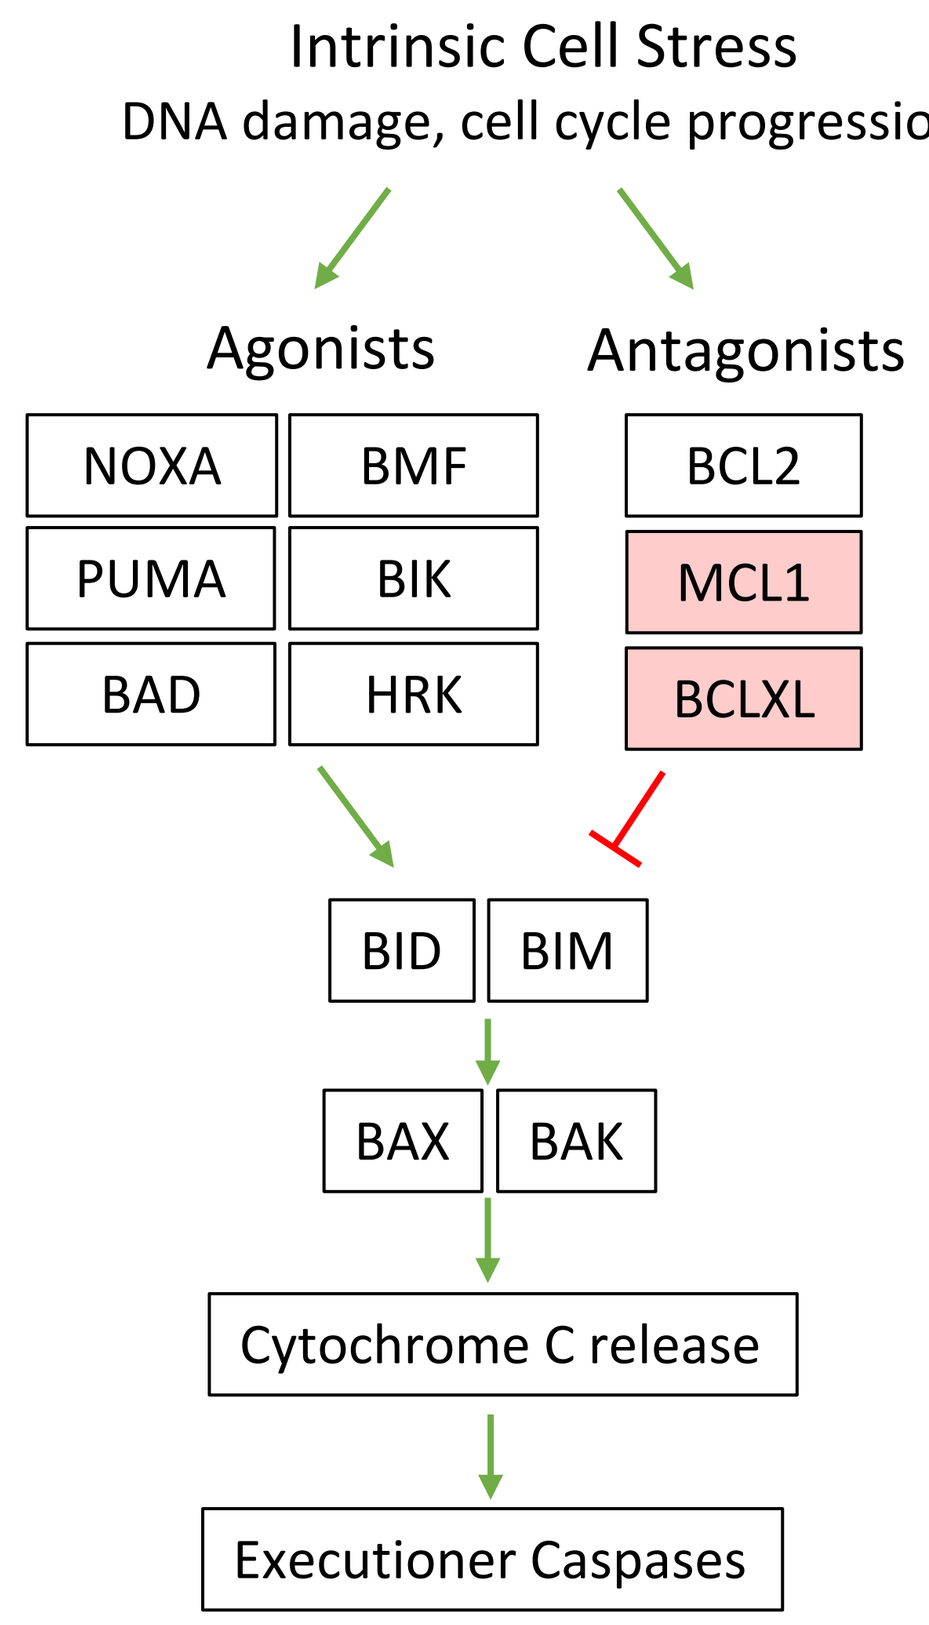

Intrinsic Cell Stress
DNA damage, cell cycle progression
Agonists
Antagonists
NOXA
BMF
PUMA
BIK
BAD
HRK
BCL2
MCL1
BCLXL
BID
BIM
BAX
BAK
Cytochrome C release
Executioner Caspases

SY-1365 induces MCL1 down-regulation in vitro and in vivo

TNBC cell lines

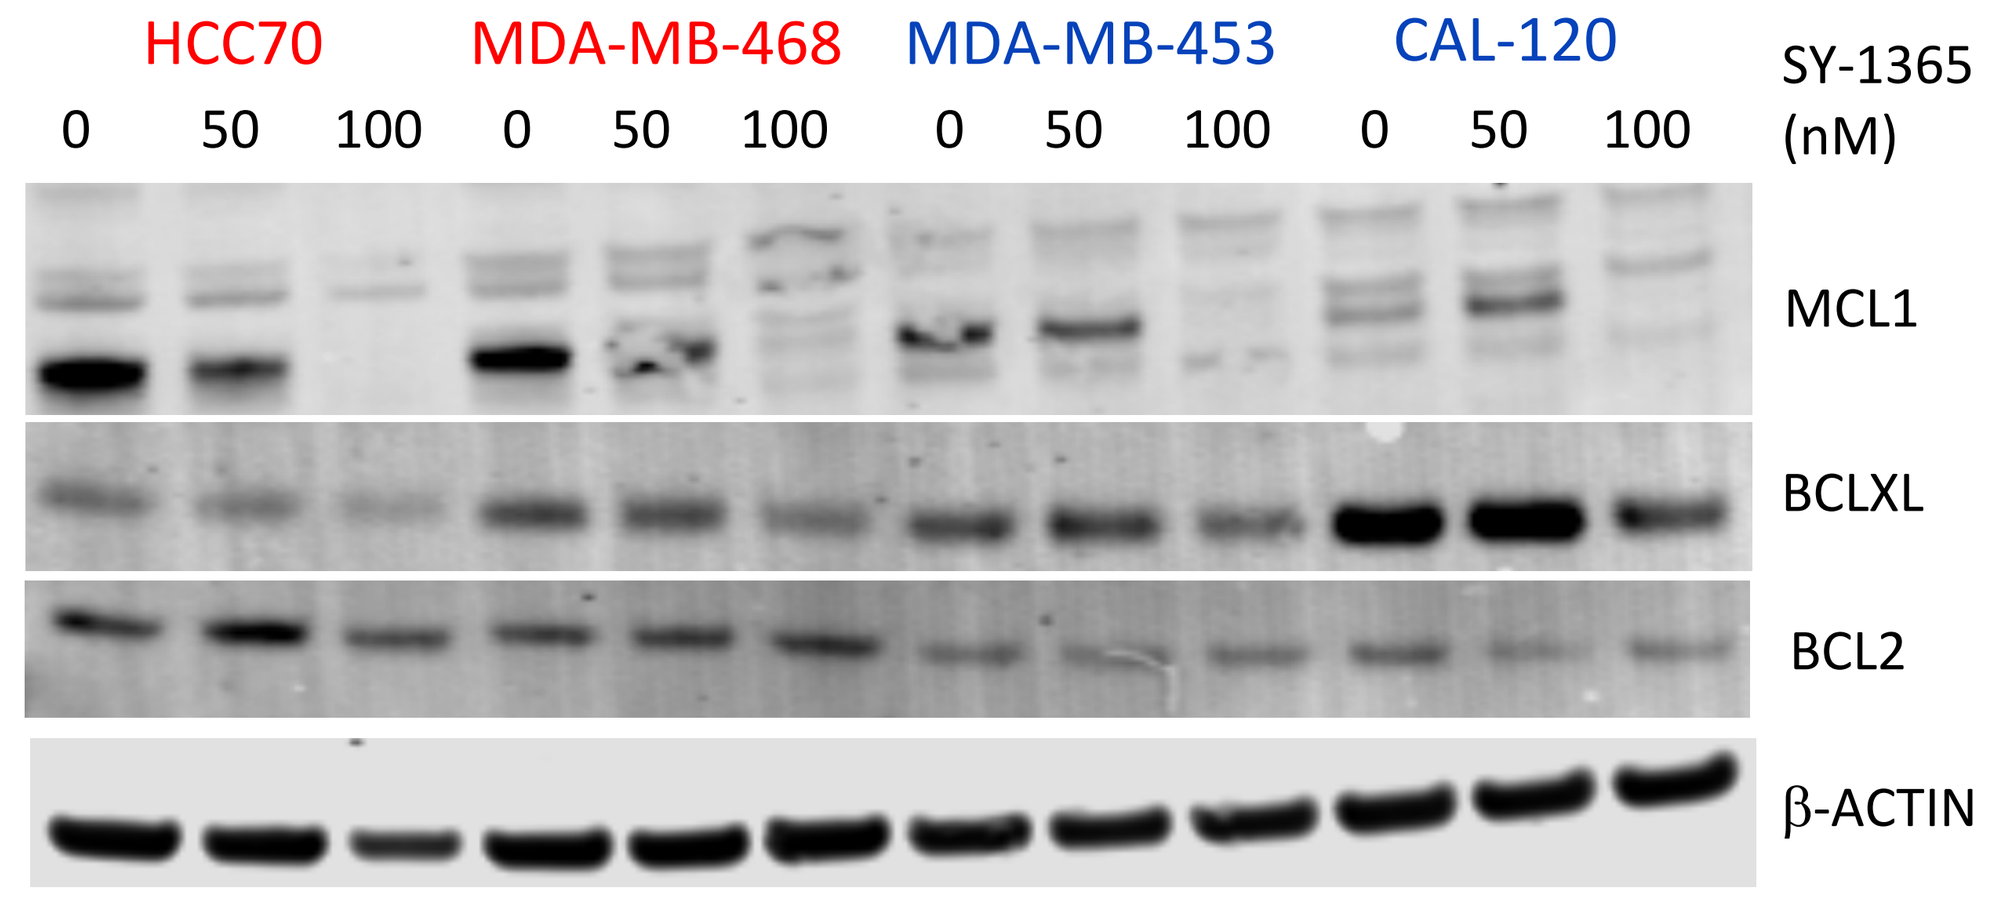

HCC70
MDA-MB-468
MDA-MB-453
CAL-120
SY-1365 (nM)
0 50 100 0 50 100 0 50 100 0 50 100
MCL1
BCLXL
BCL2
β-ACTIN

24h post SY-1365 treatment

HCC70 tumor xenografts

4h post single dose

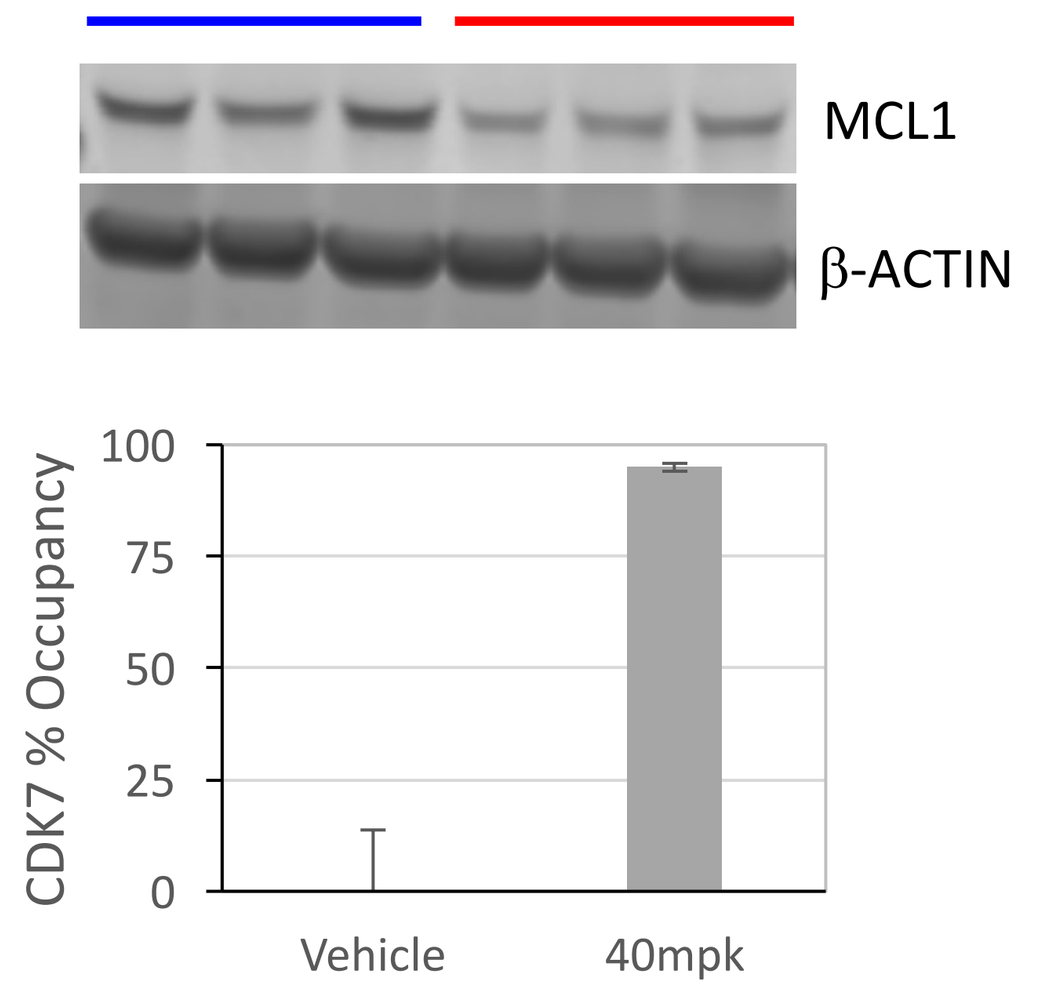

Vehicle
40 mg/kg
MCL1
β-ACTIN
CDK7 % Occupancy
Vehicle
40mpk

SY-1365 in vivo response

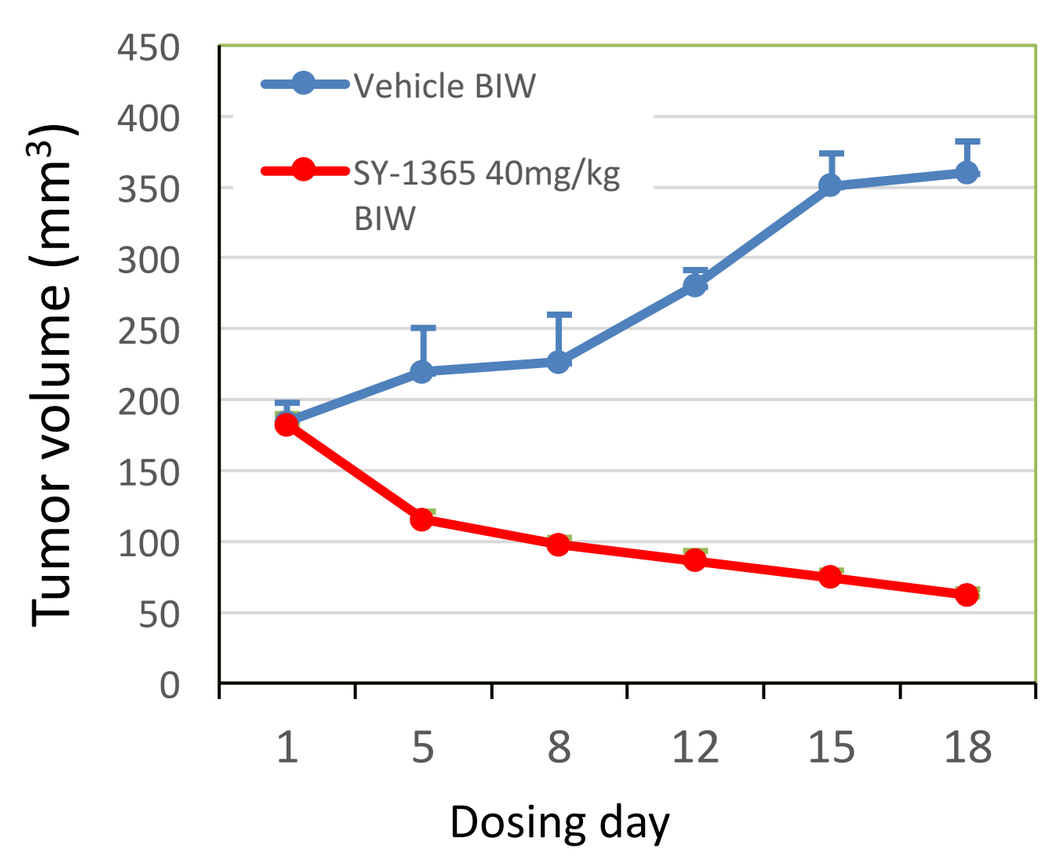

Vehicle BIW
SY-1365 40mg/kg BIW
Tumor volume (mm³)
Dosing day

Increased SY-1365 sensitivity in TNBC cell lines is associated with a Super-Enhancer (SE) upstream of *MYC*

Representative H3K27Ac ChIP-seq tracks at the *MYC* locus in SY-1365 sensitive and insensitive TNBC lines

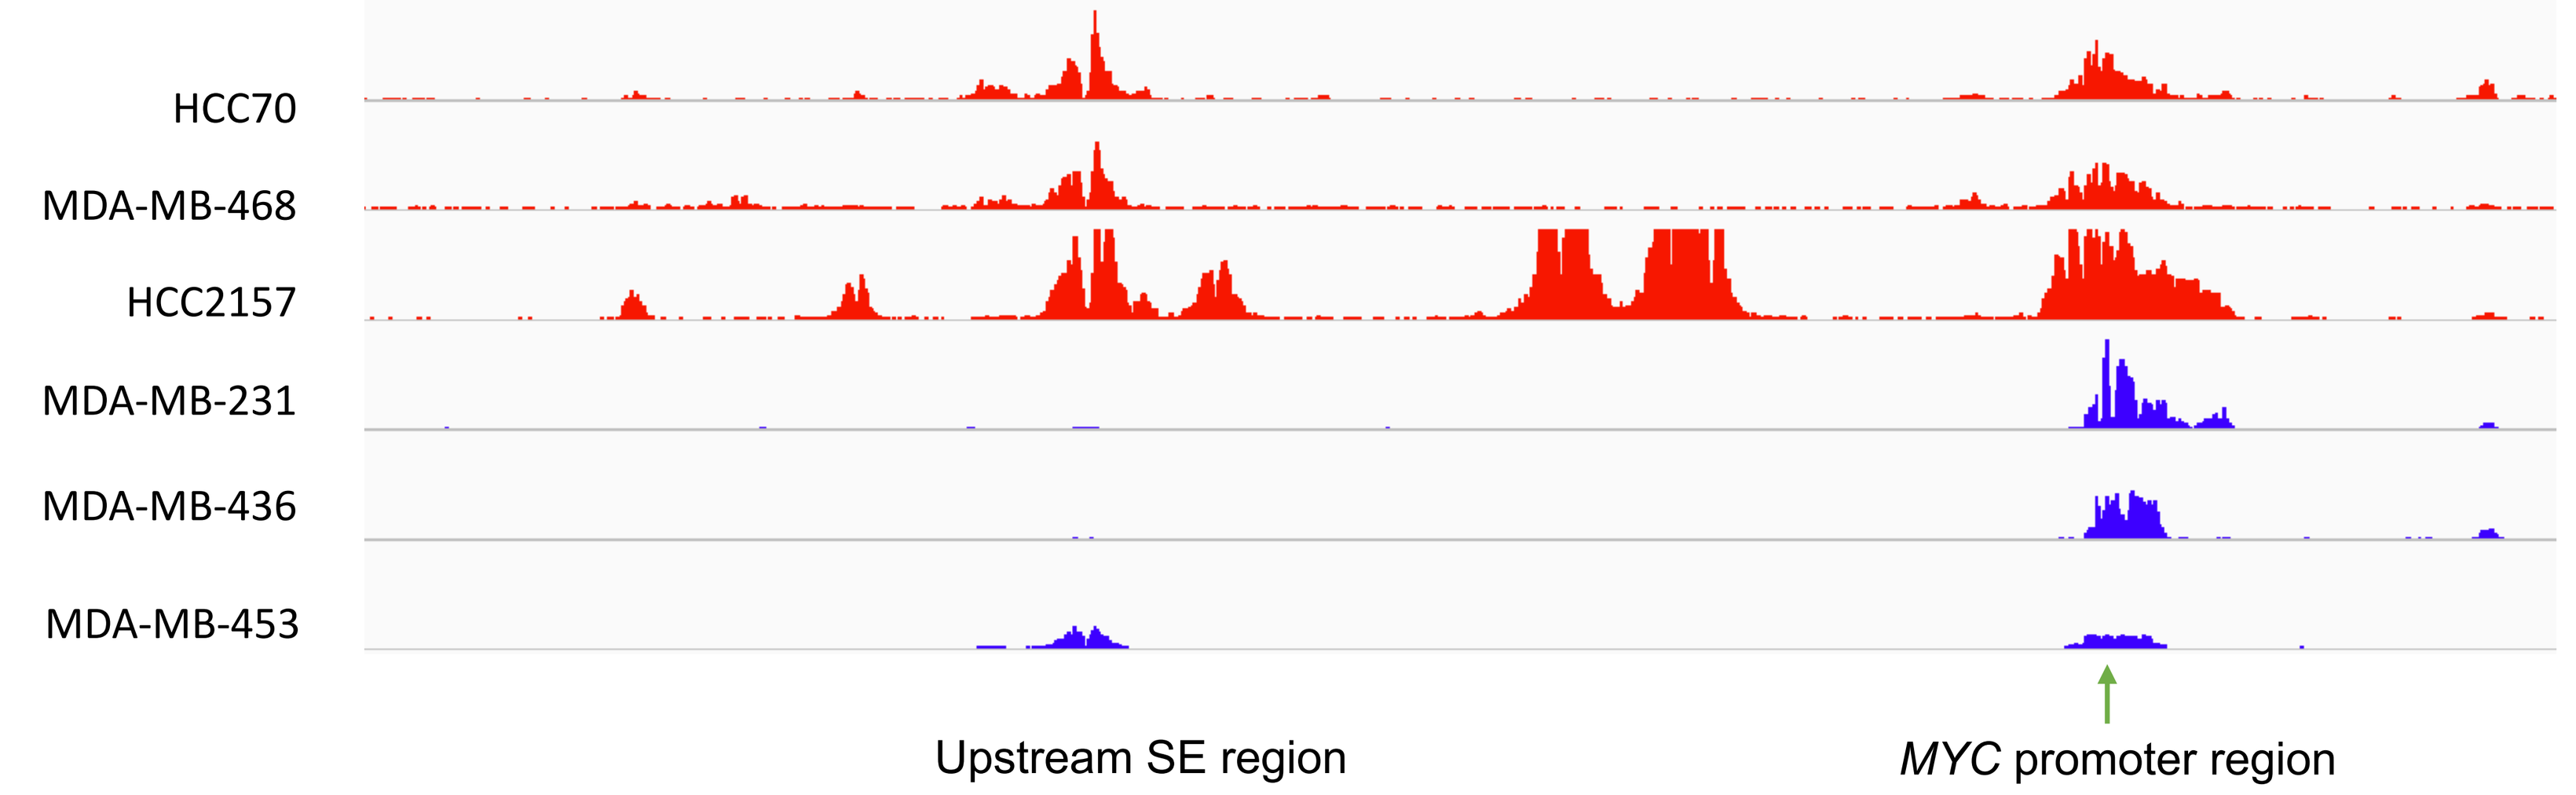

HCC70
MDA-MB-468
HCC2157
MDA-MB-231
MDA-MB-436
MDA-MB-453
Upstream SE region
MYC promoter region

Increased *MYC*-SE strength is associated with higher SY-1365 sensitivity in TNBC cells in vitro

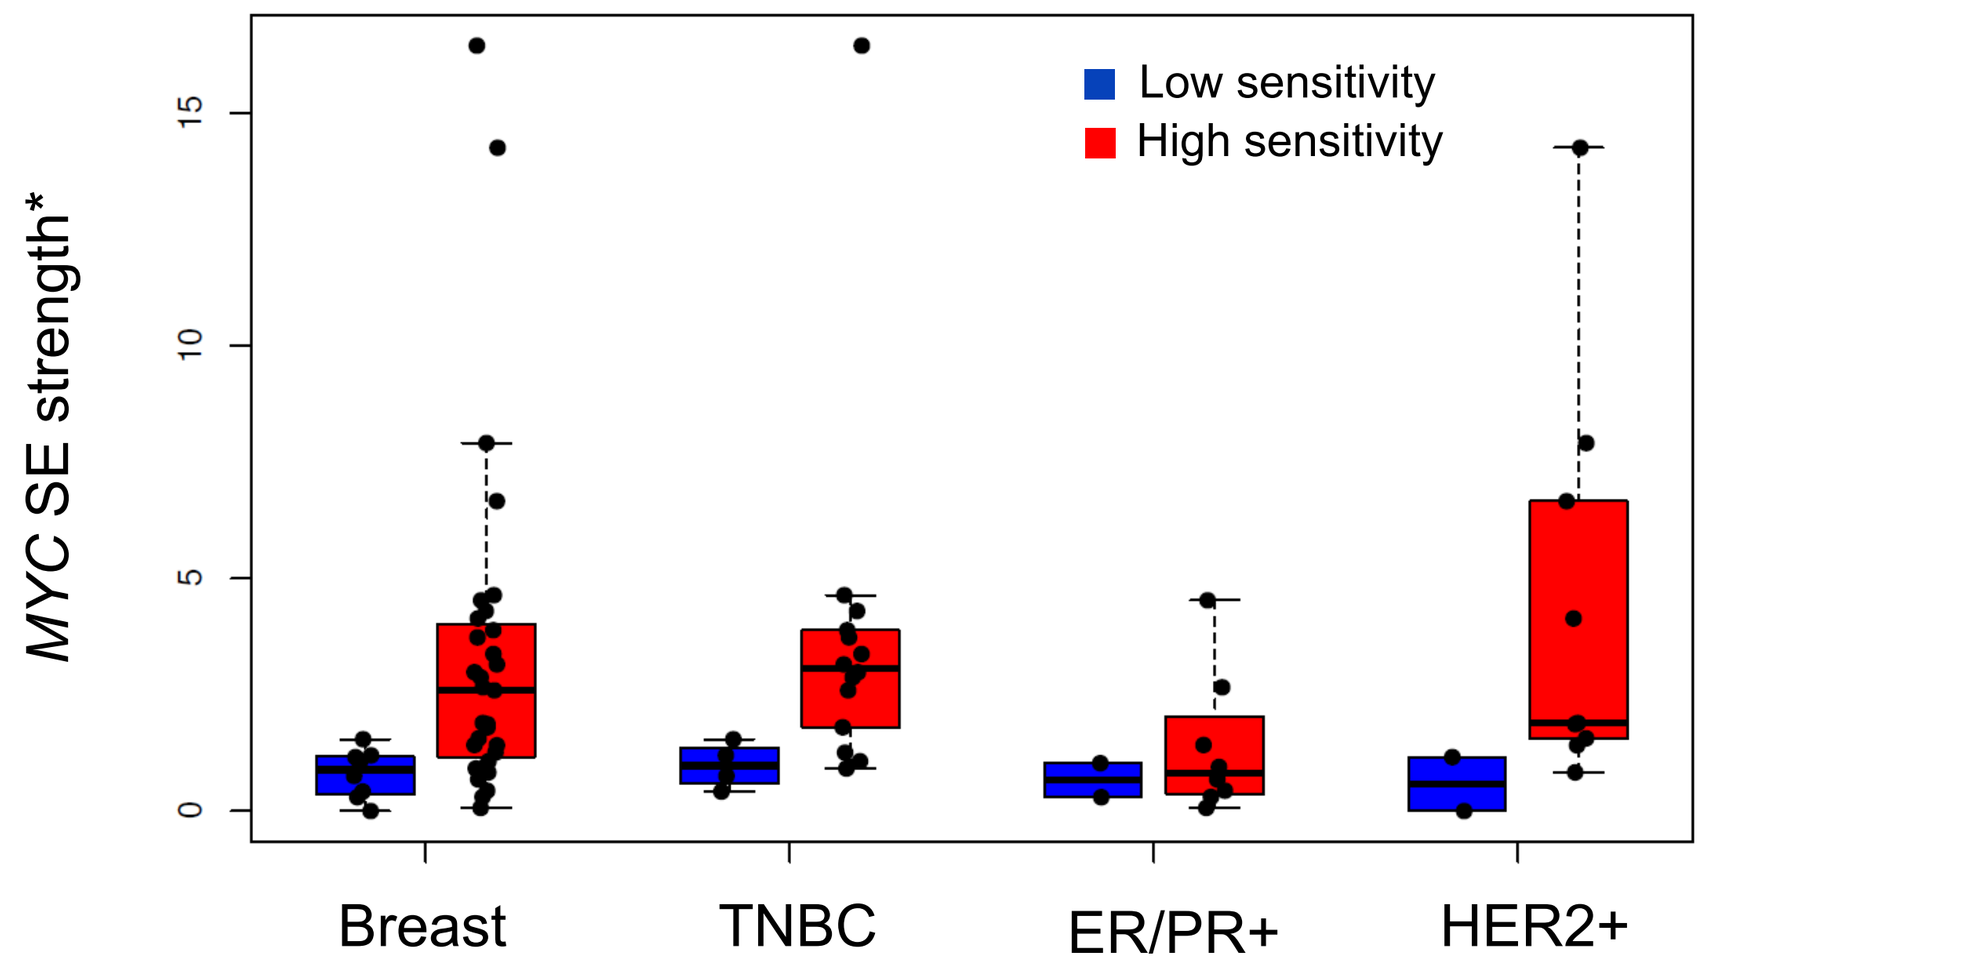

MYC SE strength*
Low sensitivity
High sensitivity
Breast
TNBC
ER/PR+
HER2+

*SE-strength is a metric based on the number of sequencing reads identified at a locus following H3K27Ac immunoprecipitation and DNA sequencing. A value of 0 means no H3K27Ac at the locus off interest; values between 0 and 1 represent typical enhancers; values >1 represent super-enhancers.

Increased *MYC*-SE strength is associated with increased *MYC* expression in TNBC cells in vitro

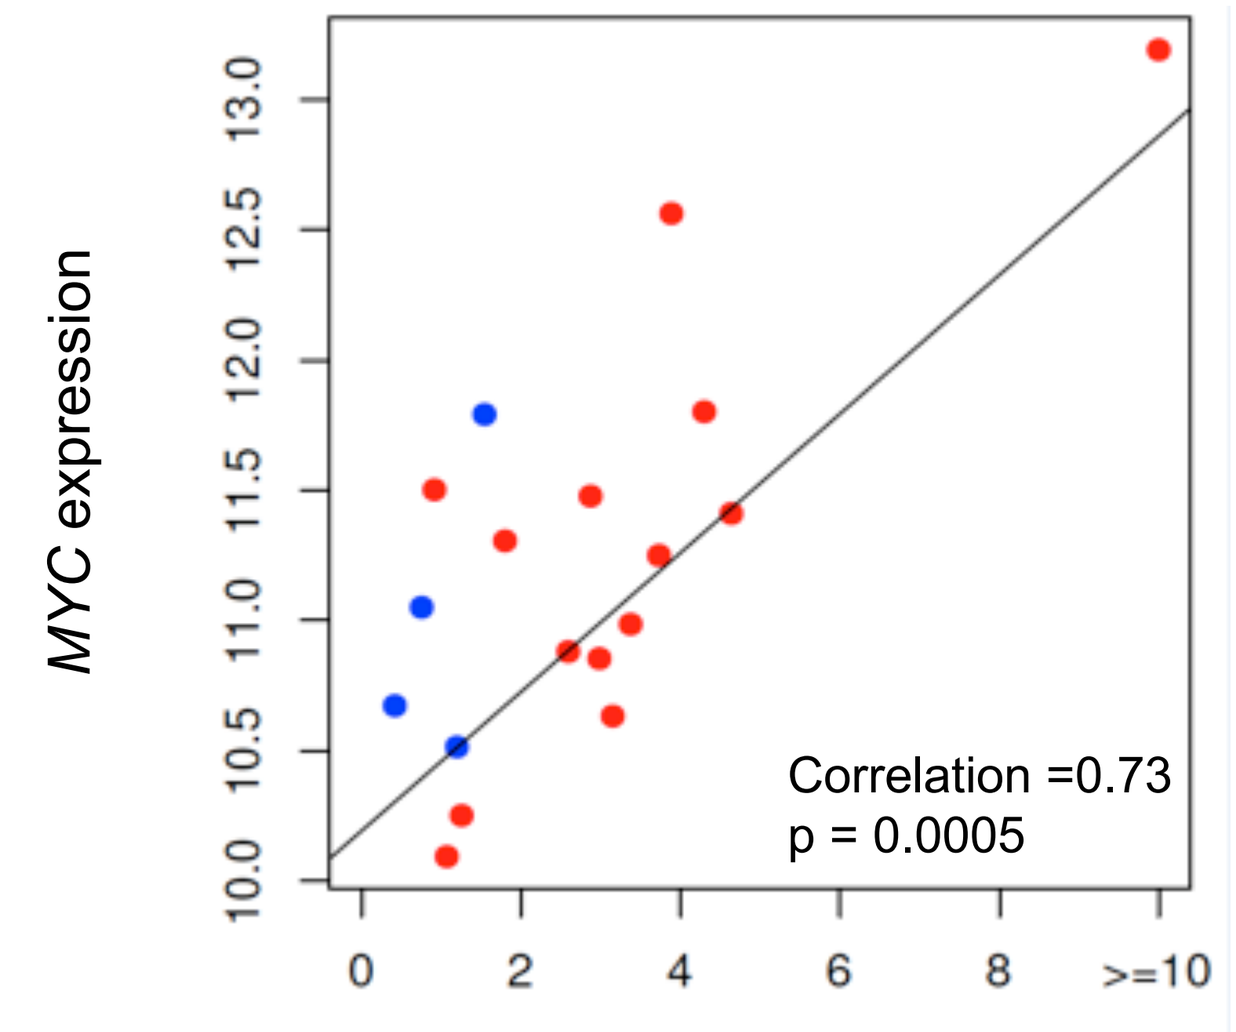

MYC expression
Correlation =0.73
p = 0.0005
MYC SE strength*

Conclusions

- SY-1365 induces cytotoxicity in breast cancer cell lines in vitro, including TNBC and ER+/PR+/HER2+ lines
- Sensitivity to SY-1365 in TNBC cell lines is associated with low expression of the mitochondrial apoptosis antagonist *BCL2L1*/BCLXL, and with increased super-enhancer strength in the transcriptional regulatory region of *MYC*
- SY-1365 induces down-regulation of the mitochondrial apoptosis antagonist MCL1 in TNBC cell lines in vitro and in HCC70 TNBC xenografts in vivo
- Down-regulation of MCL1 in HCC70 xenografts is associated with SY-1365 target engagement (CDK7 occupancy), and tumor regressions with an intermittent dosing regimen
- These results highlight the potential for SY-1365 in the treatment of TNBC. Further exploration of SY-1365 activity and development of predictive markers of response in TNBC are warranted
- SY-1365 is currently being assessed in a phase 1 trial in adult patients with advanced solid tumors (NCT03134638).

San Antonio Breast Cancer Symposium, San Antonio TX. Dec 5-9, 2017