Safety of BXCL701, a small molecule inhibitor of dipeptidyl peptidases 8/9 (DPP), with pembrolizumab, (pembro, anti-PD-1) monoclonal antibody, in men with metastatic castration-resistant prostate cancer (mCRPC)

Rahul R. Aggarwal[1], Dan Costin[2], Vincent O'Neill[3], Cedric Burg[3], Diane Healey[3], Johann S. de Bono[4], Jingsong Zhang[5], Eric Jay Small[1]

1. UCSF Helen Diller Family Comprehensive Cancer Center, San Francisco, CA; 2. Center for Cancer Care at White Plains Hospital, White Plains, NY; 3. BioXcel Therapeutics, New Haven, CT; 4. Royal Marsden NHS Foundation Trust, London, United Kingdom; 5. H. Lee Moffitt Cancer Center & Research Institute, Tampa, FL

Abstract No- 140

BACKGROUND

- BXCL701 (talabostat) is an orally administered systemic activator of the innate immunity pathway and inhibitor of immune evasion via inhibition of DPP8/9 (dipeptidyl peptidases) and FAP (fibroblast activation protein)
- BXCL701 has an established tolerable safety profile (>700 subjects)
 - Manageable and usually transient side effects consistent with cytokine activation
 - BXCL701 single agent anti-tumor activity seen in metastatic melanoma
 - Prior development program (2000-2007) focused on chemo- combinations
- BXCL701 current program is focused on combination therapies with newer immuno-oncology treatments, such as anti-PD(L)-1 agents

BXCL701 MECHANISM OF ACTION

BXCL701 Therapeutic Potential in Solid Tumors: Response of cancer to IO treatment relies on a fully active immune response

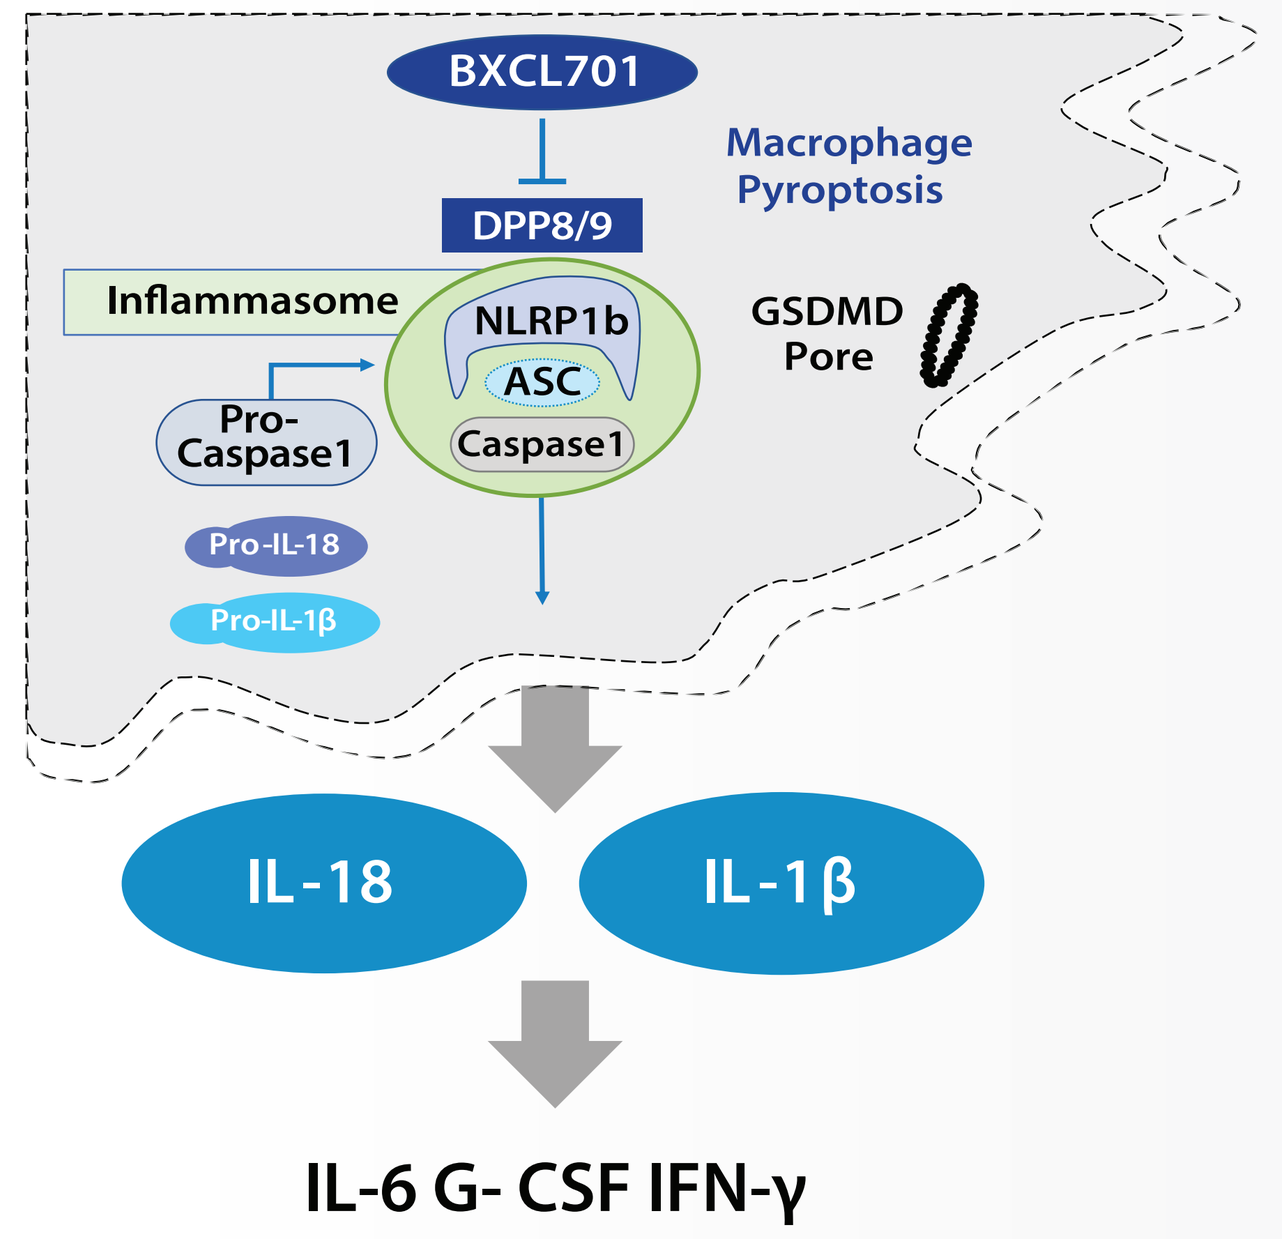
BXCL701
DPP8/9
Macrophage Pyroptosis
Inflammasome
NLRP1b
ASC
Caspase1
Pro-Caspase1
GSDMD Pore
Pro-IL-18
Pro-IL-1β
IL-18
IL-1β
IL-6 G- CSF IFN-γ

- Inhibition of DPP8/9 causes an **innate immunogenic reaction** in the macrophages called **pyroptosis**
- Pyroptosis is a pro-inflammatory pathway to cell death and drives the activation of caspase-1 and subsequent activation of pro-IL-1β and pro-IL-18 leading to the **production of a host of other cytokines and chemokines including IL-1β & IL-18**
- The release of these cytokines and chemokines may result in increased sensitivity and **synergistic anti-tumor activity when combined** with other agents that target the **adaptive immunogenic response** such as checkpoint inhibitors which largely target T-cells.

BXCL701 TARGET AMPLIFICATION IN SMALL - CELL/ NEUROENDOCRINE PROSTATE CANCER

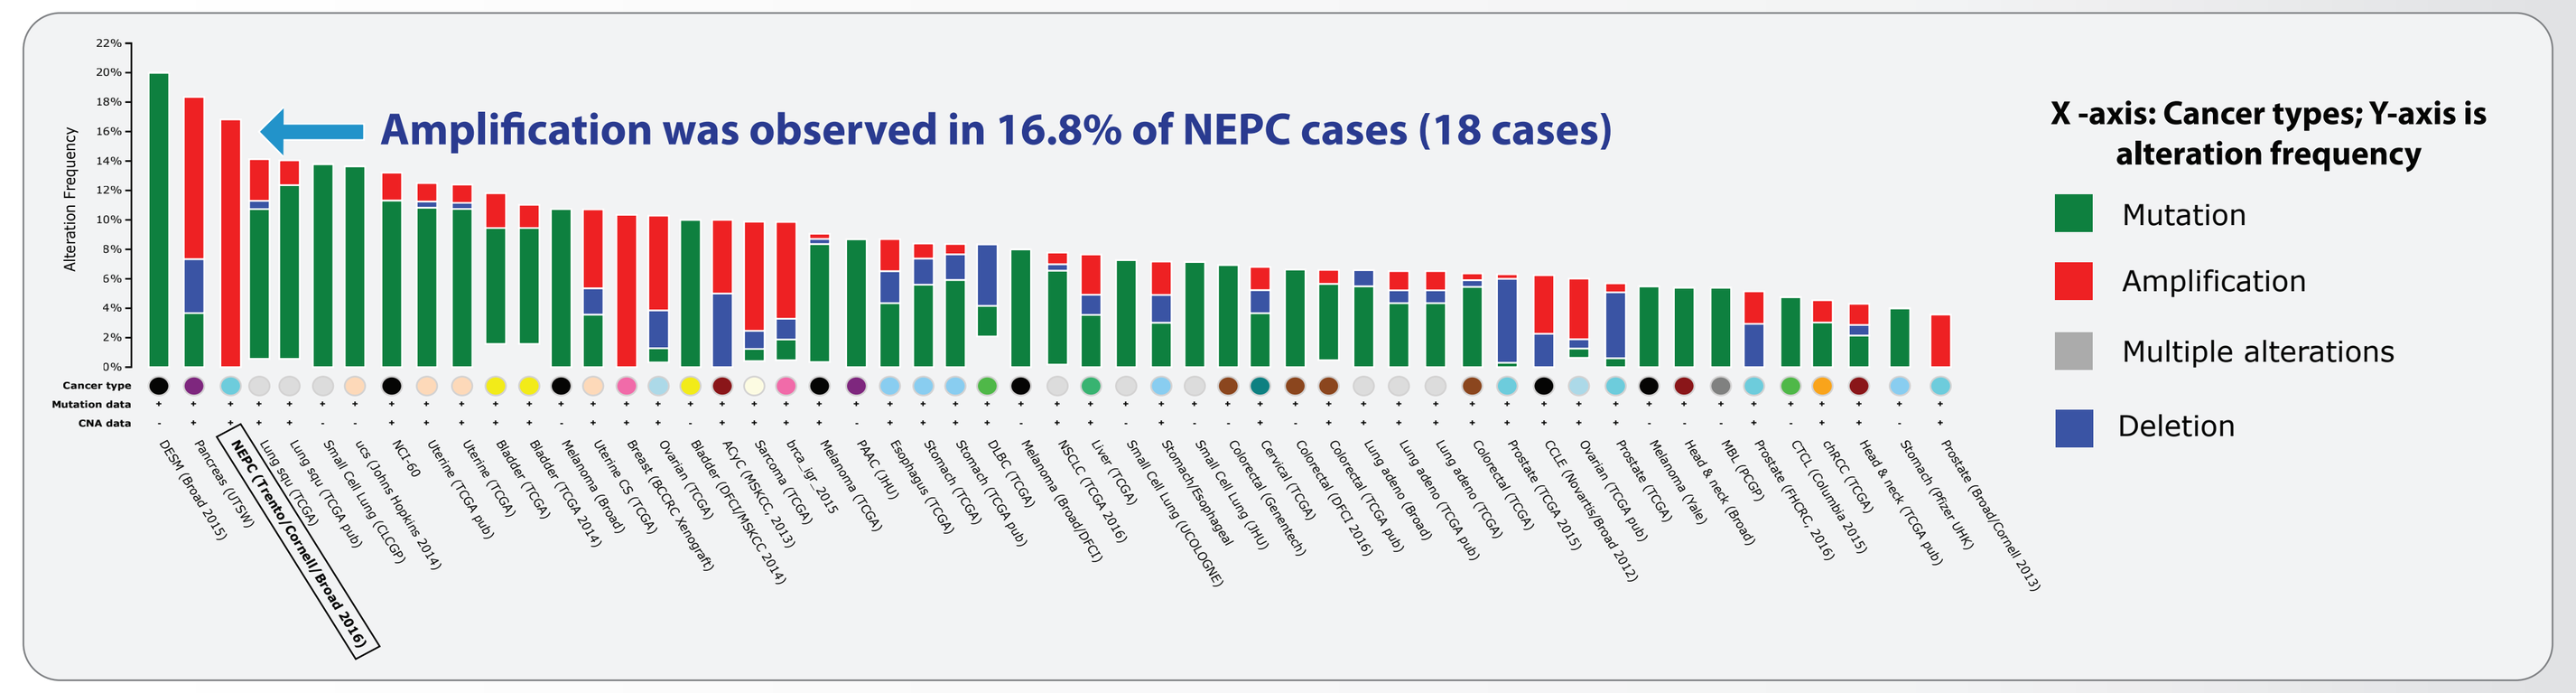
Amplification was observed in 16.8% of NEPC cases (18 cases)
X -axis: Cancer types; Y-axis is alteration frequency
Mutation
Amplification
Multiple alterations
Deletion
Alteration Frequency

An analysis of genomic alterations in DPP8 and DPP9 across cancer genomic profiles of 33172 patients from 150 different cancer studies included dataset from TCGA

CO-AMPLIFICATION AND OVER EXPRESSION OF BXCL701 TARGETS WITH PDL1 (CD247)

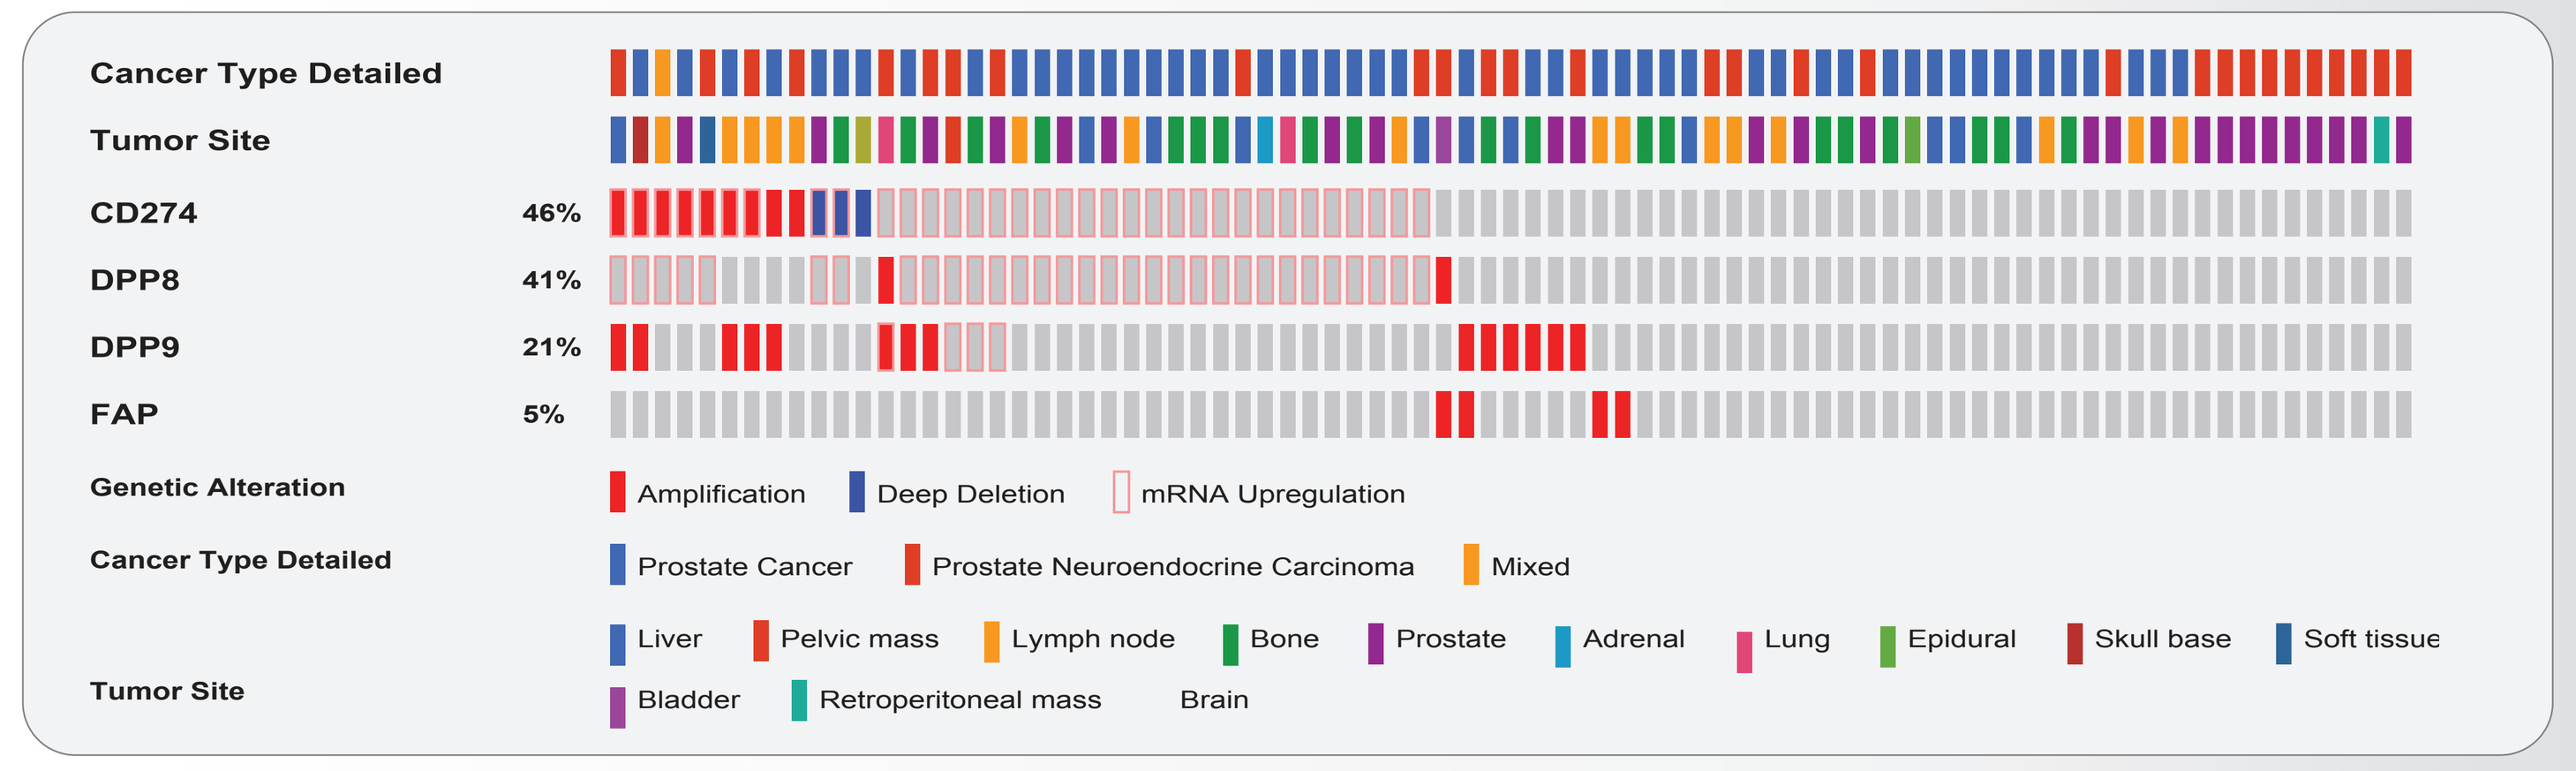
Cancer Type Detailed
Tumor Site
CD274 46%
DPP8 41%
DPP9 21%
FAP 5%
Genetic Alteration
Amplification
Deep Deletion
mRNA Upregulation
Cancer Type Detailed
Prostate Cancer
Prostate Neuroendocrine Carcinoma
Mixed
Tumor Site
Liver
Pelvic mass
Lymph node
Bone
Prostate
Adrenal
Lung
Epidural
Skull base
Soft tissue
Bladder
Retroperitoneal mass
Brain

PRECLINICAL DATA VALIDATES BXCL701 AND ANTI-PD1 COMBINATION POTENTIAL

Treatment Effect Of BXCL701 With Anti-pd-1 On Mean Tumor Volume

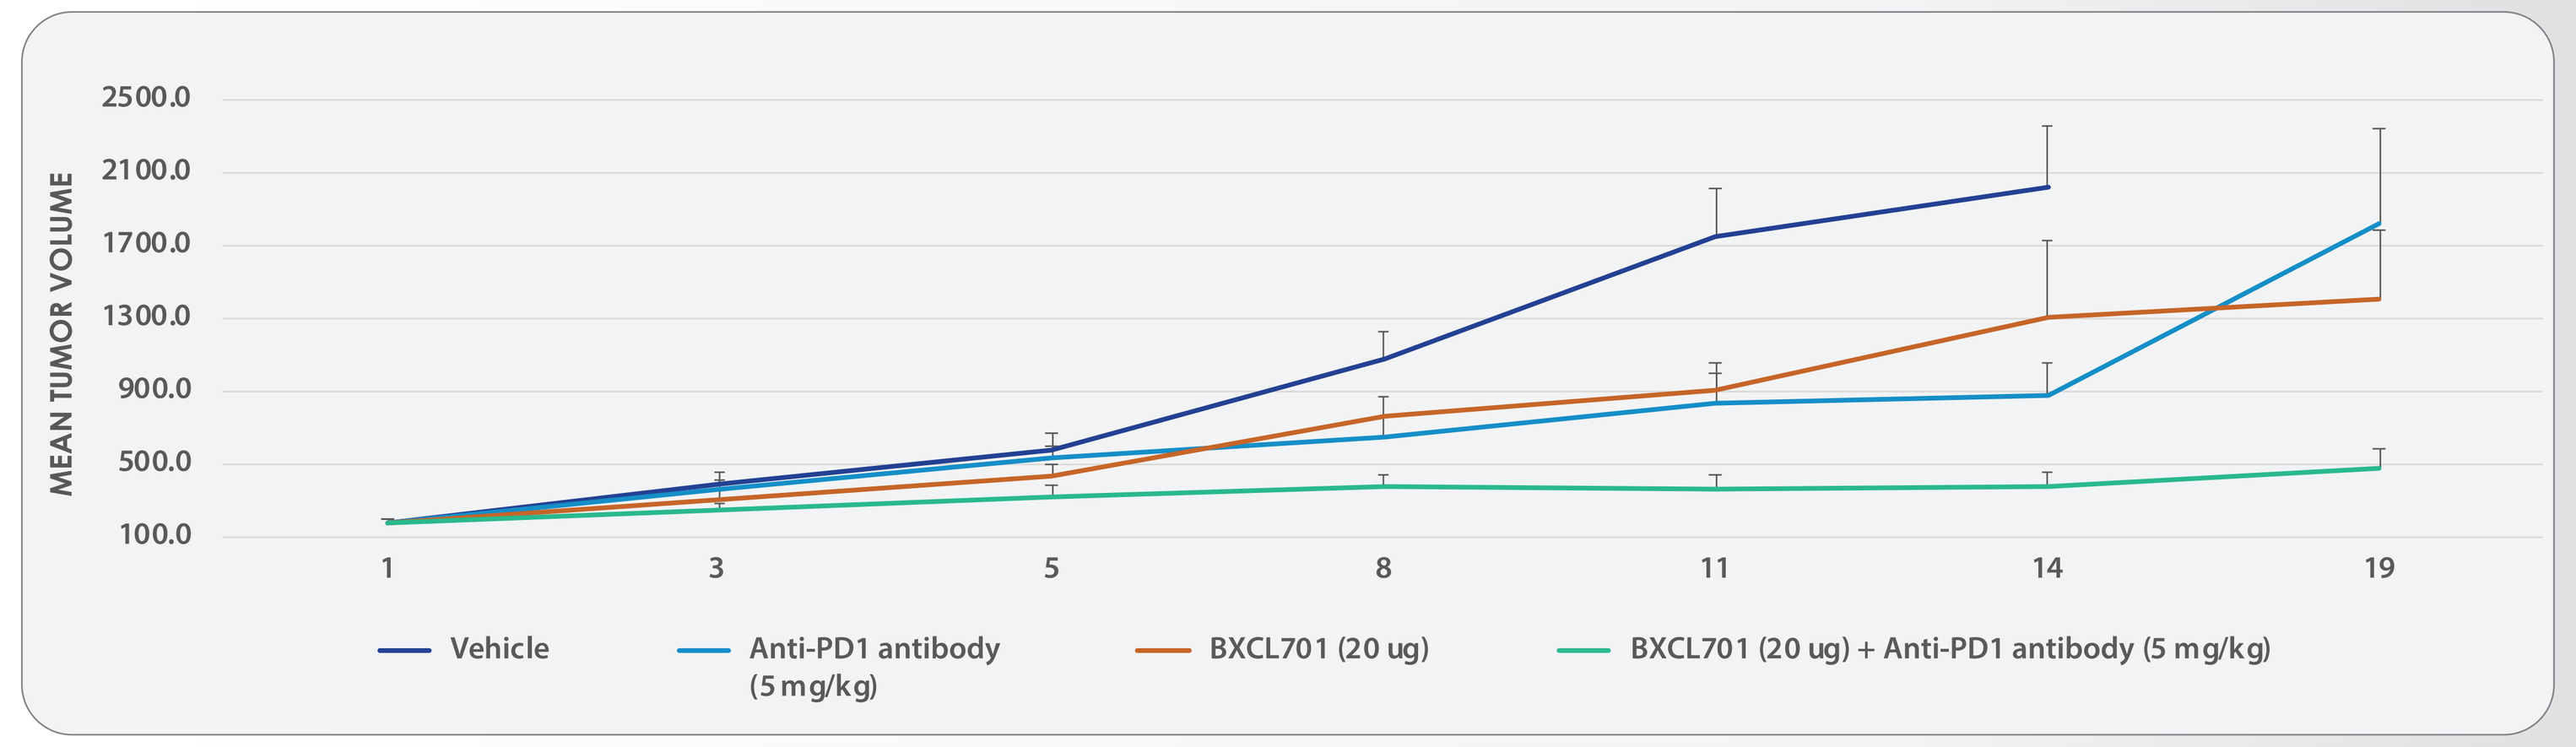
MEAN TUMOR VOLUME
2500.0
2100.0
1700.0
1300.0
900.0
500.0
100.0
1
3
5
8
11
14
19
Vehicle
Anti-PD1 antibody (5 mg/kg)
BXCL701 (20 ug)
BXCL701 (20 ug) + Anti-PD1 antibody (5 mg/kg)

- Synergistic antitumor effect on **tumor volume** in MC38 mouse model of colon adenocarcinoma
- Synergistic **upregulation in the immunomodulatory parameters** for pro-Inflammatory cytokines*, chemokines⁺ and associated memory T-cells# by dose as single agent and in combination
- Synergistic **increase in the cytotoxic NK cells and macrophages** in the tumor with a **decrease in the immunosuppressive T-regs**

***IL-2, IL-12p40, IL-6; +GM-CSF, G-CSF; #IL-15, IL-7**

AACR2017 Abstract 2629

DE-NOVO OR TREATMENT-EMERGENT SMALL-CELL / NEUROENDOCRINE PROSTATE CANCER: UNMET TREATMENT NEED

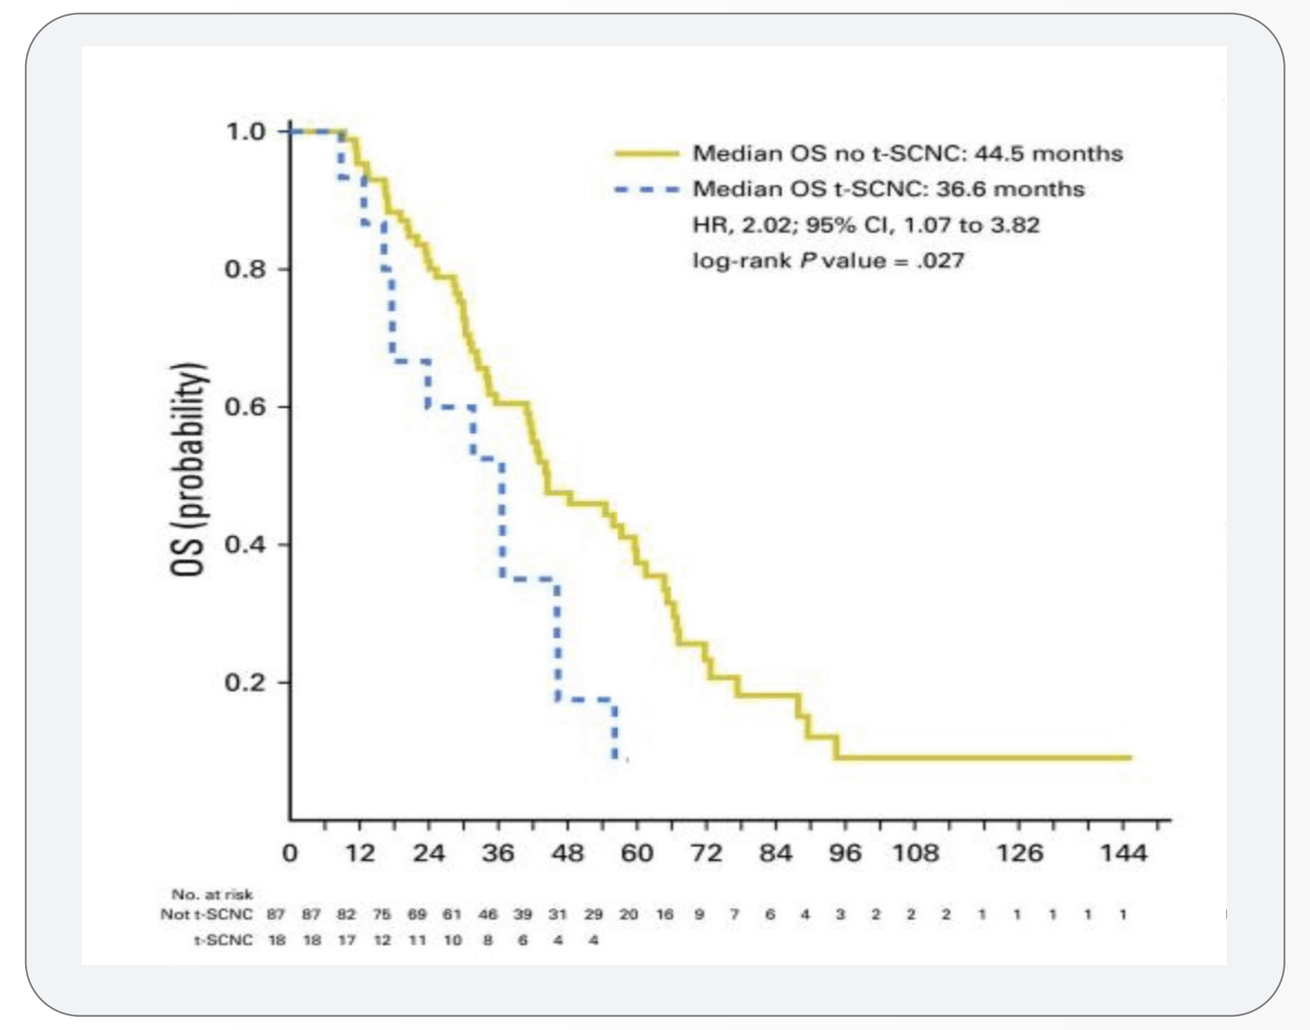
Median OS no t-SCNC: 44.5 months
Median OS t-SCNC: 36.6 months
HR, 2.02; 95% CI, 1.07 to 3.82
log-rank P value = .027
OS (probability)
1.0
0.8
0.6
0.4
0.2
0
12
24
36
48
60
72
84
96
108
126
144

- De-Novo small-cell cancer of the prostate is present in <1% of newly diagnosed cases, associated with AR-null phenotype and progression with low serum PSA levels
- Treatment-emergent SC/NEPC may reflect a trans-differentiation process after androgen -ablating therapy
- Both de-novo and treatment- emergent SC/NEPC are highly aggressive forms of prostate cancer with no current standard of care

Aggarwal R, et al. JCO 2018

PHASE 1B/2 TRIAL SCHEMATIC

SAFETY LEAD-IN: Standard 3X3 dose escalation with expansion dependent on DLTs.

→ **BXCL701 0.4 mg/ qd + Pembro (n= 3) Completed**

BXCL701 0.6 mg/qd + Pembro (n=3) Completed

BXCL701 0.6mg split dose/qd + Pembro (n=6) Enrolling

Expansion Cohort

EFFICACY STAGE: Simon 2 stage Power: 0.8, Type 1 error:0.05; P0:15%, P1:35%

→ **BXCL701 @ RP2D + Pembro (n=15)** → **>2 Composite Responses** → **BXCL701 @ RP2D + Pembro (n=13)**

TREATMENT CYCLE: BXCL701 PO days 1-14 of 21 days pembrolizumab 200mg IV Q21 days

KEY INCLUSION AND EXCLUSION CRITERIA

KEY INCLUSION CRITERIA

- Patient has progressive, metastatic castration -resistant disease, as defined by PCWG3 criteria
- Progression during or following completion of at least 1 prior line of systemic therapy for locally advanced or metastatic prostate cancer
- Efficacy Stage only:
 - Evidence of SCNC on central pathology review of archival tumor tissue
 - Received at least 1 prior line of chemotherapy
 - Willing to undergo metastatic tumor biopsy during Screening.
- Serum testosterone <50 ng/dL during Screening except for those with de novo small cell prostate cancer
- Eastern Cooperative Oncology Group (ECOG) performance status of 0-2

KEY EXCLUSION CRITERIA

- Has received treatment with >2 cytotoxic chemotherapy regimens for CRPC
- Prior treatment with an anti-PD-1, anti-PD-L1, anti- programmed death- ligand 2 (PD-L2) agent or with an agent directed to another co-inhibitory T-cell receptor
- Additional active malignancy that may confound the assessment of the study endpoints
- Brain metastases that are symptomatic and progressive on imaging
- Significant cardiovascular or pulmonary disease

SAFETY LEAD-IN RESULTS (30JAN20 DATA-CUT)

PATIENT DISEASE HISTORY

DISEASE HISTORY		NO. OF PATIENTS BXCL701 0.4mg + Pembro (N=3)	NO. OF PATIENTS BXCL701 0.6mg + Pembro (N=3)
Most Recent Histopathology	Adenocarcinoma	3	2
	Primary Small cell neuroendocrine	-	-
	Mixed adenocarcinoma + SC/NEPC	-	1
Prior Systemic Therapies	ADT therapy	3	3
	Chemotherapy	3	1
	Radiotherapy	2	3
Type of Progression at Study Entry	PSA only	1	-
	Bone +/- PSA	1	1
	Nodal disease + PSA	-	1
	Bone + nodal disease +/- PSA	1	1
	Visceral +/- other	-	-

EXPOSURE DURATION AND SUBJECT DISPOSITION

PATIENT #	C1	C2	C3	C4	C5	C6	C7	C8	C9	C10	OFF TX REASON
Cohort #1 BXCL701 0.4mg + Pembro, N=3											
#1	X	X	X	X	X	X	DC				Progression
#2	X	X	X	X	X	X	X	X	X	X	
#3	X	X	X	X	X	X	X	X	X		
Cohort #2 BXCL701 0.6mg + Pembro N=3											
#4	X	X	X	DC							Adverse Event
#5	X	X	X	X							
#6	X	X	X	X							

C= Cycle defined as 21 days: BXCL701 PO days 1-14 + pembrolizumab 200mg IV day 1

SAFETY SUMMARY

AE CATEGORY	NO. OF PATIENTS BXCL701 0.4mg + Pembro N=3	NO. OF PATIENTS BXCL701 0.6mg + Pembro N=3
Any DLT	0	1
Any treatment-related SAE*	0	1
Any BXCL701 treatment-related AE*	3	3
Any pembro treatment-related AE*	3	2
Any BXCL701 or pembro treatment-related Grade 3 or 4 AE*		
Non-hematologic	-	2***
Hematologic	2**	-

*Includes related and possibly-related events | ** 2pts: Grade 3 thrombocytopenia | *** 1pt each: Grade 3 syncope (DLT C1D6) and acidosis with fatal outcome (C3)

AE TYPE: System Organ Class	Preferred Term	BXCL701 0.4mg + Pembro N=3 ALL CAUSE	BXCL701 0.4mg + Pembro N=3 RELATED	BXCL701 0.6mg + Pembro N=3 ALL CAUSE	BXCL701 0.6mg + Pembro N=3 RELATED
AEs of Special Interest					
Vascular Disorders	Hypotension*	-	-	3	3
Nervous System Disorders	Dizziness	-	-	2	2
	Syncope	-	-	1	1
Respiratory, Thoracic and Mediastinal Disorders	Dyspnea	-	-	2	1
General Disorders	Chills	-	-	-	-
	Edema	1	1	2	2
Musculoskeletal and Connective Tissue	Myalgia	-	-	1	1
Other AEs in ≥ 2 pts					
Blood /Lymphatic Disorders	Anemia	3	2	-	-
Gastrointestinal Disorders	Constipation	-	-	2	-
General Disorders	Fatigue/Malaise	1	1	1	1
Metabolism and Nutrition Disorders	Hypocalcemia	2	1	1	1

* Includes orthostasis and orthostatic hypotension

EFFICACY

ASSESSMENT	BXCL701 0.4mg qd + PEMBRO N=3	BXCL701 0.6mg qd + PEMBRO N=3
RECIST 1.1 Response		
SD	2	1
Non-CR/Non-PD	1*	1*
NE	-	1**
PSA decline >50% by week 12	0	0

**bone only mets at baseline*
*** 1pt with PSA only disease reported*

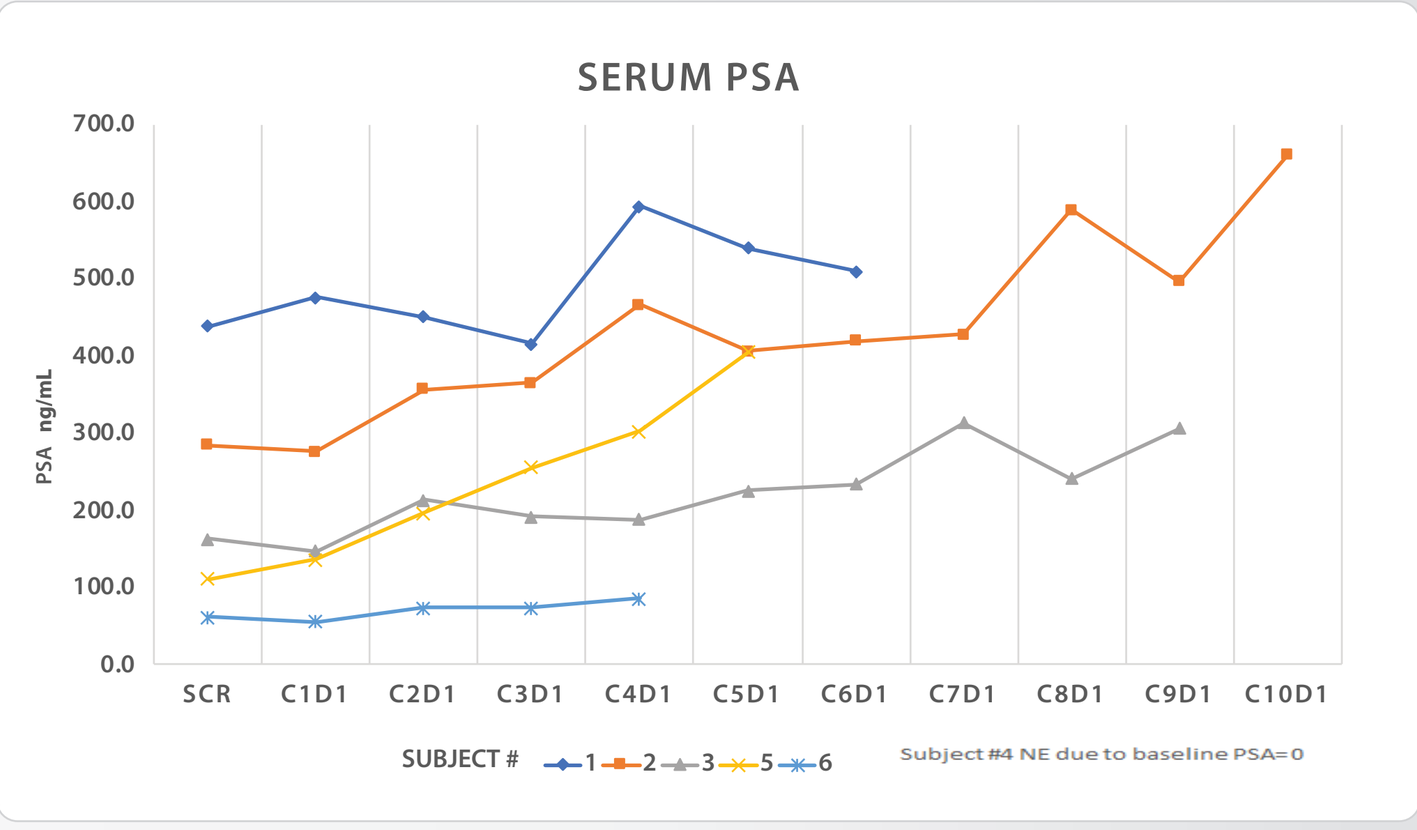
SERUM PSA
PSA ng/mL
700.0
600.0
500.0
400.0
300.0
200.0
100.0
0.0
SCR
C1D1
C2D1
C3D1
C4D1
C5D1
C6D1
C7D1
C8D1
C9D1
C10D1
SUBJECT # 1 2 3 5 6
Subject #4 NE due to baseline PSA=0

CONCLUSIONS

- BXCL701 is an orally available innate immune activator and inhibitor of immune evasion
- In the Phase 1b safety lead-in portion of this study in subjects with mCRPC, BXCL701 + pembro:
 - Has demonstrated on-target side effects consistent with cytokine activation at the highest once-daily dose tested
 - Preliminary pharmacokinetics of BXCL701 are within expectations based on prior data
 - The majority of patients remain on treatment
- In order to improve tolerance, bid dosing is being explored in the expansion cohort.
- The Phase 2 portion of this study will be limited SC/NEPC patients and will assess the anti-tumor activity of the combination in a setting where checkpoint inhibitor monotherapies have demonstrated limited clinical benefit

ACKNOWLEDGMENTS

All patients, their families, and caregivers who make this study possible; the participating investigators and their staff; Iqvia Biotech for assisting in the conduct of the study and S. Corsi-Travali and J. MacDougall of BioXcel Therapeutics.

American Society of Clinical Oncology Genitourinary Cancers Symposium- February 13-15, 2020, San Francisco, CA