Phase 1b/2 study of BXCL701, an oral activator of the systemic innate immunity pathway, combined with pembrolizumab (pembro), in men with metastatic castration-resistant prostate cancer (mCRPC)

Abstract ID: 341

Rahul Aggarwal[1], MD, Dan Costin, MD[2], Jingsong Zhang, MD, PhD[3], Paul Monk, MD[4], Mark Linch, MD[5], Lawrence I. Karsh, MD[6], Diane Healey, MS[7], Stefani Corsi-Travali[7], Sreenivas Adurthi, PhD[7], Adedayo Adedoyin[7], Vincent O'Neill, MD[7]

1. UCSF Helen Diller Family Comprehensive Cancer Center, 2. White Plains Hospital, Center for Cancer Care, 3. H. Lee Moffitt Cancer Center and Research Institute, 4. The Ohio State University Comprehensive Cancer Center 5. University College London Cancer Institute, 6. The Urology Center of Colorado, 7. BioXcel Therapeutics

METASTATIC CASTRATION-RESISTANT PROSTATE CANCER (mCRPC) UNMET MEDICAL NEED

- American Cancer Society estimates ~191,930 new cases of prostate cancer in U.S. for 2020, 10–20% develop CRPC within approximately 5 years of follow-up, most of them having metastases at time of diagnosis
- Treatment of mCRPC has evolved rapidly over the past few years: first line treatment with androgen deprivation therapy (ADT) or one of the newer androgen signaling inhibitors (ASI), abiraterone or enzalutamide followed by chemotherapy with docetaxel is now standard of care. Docetaxel is associated with a median overall survival of less than 2 years
- Treatment Emergent Neuroendocrine Prostate Cancer (t-NEPC) is an aggressive tumor that appears in about 20% of patients progressing after treatment with ADT or ASI. This phenotype is typically treated with chemotherapy and associated with a shortened survival compared to the more common adenocarcinoma phenotype
- Despite demonstrated benefit of immunotherapies targeting PD-1—such as pembrolizumab—on clinical outcomes in many solid tumors, mCRPC remains largely resistant to such therapies with single agent response rates around 6% (Keynote 199 ASCO-GU 2020). Further exploration has been focused on combination therapies
- The BXCL701 immunomodulatory mechanism may manipulate the tumor micro-environment in such a way turning a "cold" tumor environment into an inflamed "hot" environment so that prostate cancer can overcome resistance to immunotherapy

BXCL701 MECHANISM OF ACTION

- A critical role of the innate immune system is to alert the adaptive immune system of an invading pathogen
 - Induction of Inflammation mediated through cytokines
 - Stimulation of antigen presentation by professional antigen presenting cells
- BXCL701 (Talabostat/Val-BoroPro) is a first-in-class oral small molecule inhibitor of dipeptidyl peptidases (DPP) DPP8 and DPP9
- BXCL701 is designed to trigger inflammasome mediated pyroptosis to alert and prime adaptive immune cells, leading to induction of IL-18 and IL-1ß, bridging between innate and adaptive immunity
- Activation of innate immunity has been shown to complement T-cells targeting immunotherapy: in syngeneic animal models, significant tumor responses were observed with BXCL701 plus checkpoint inhibition

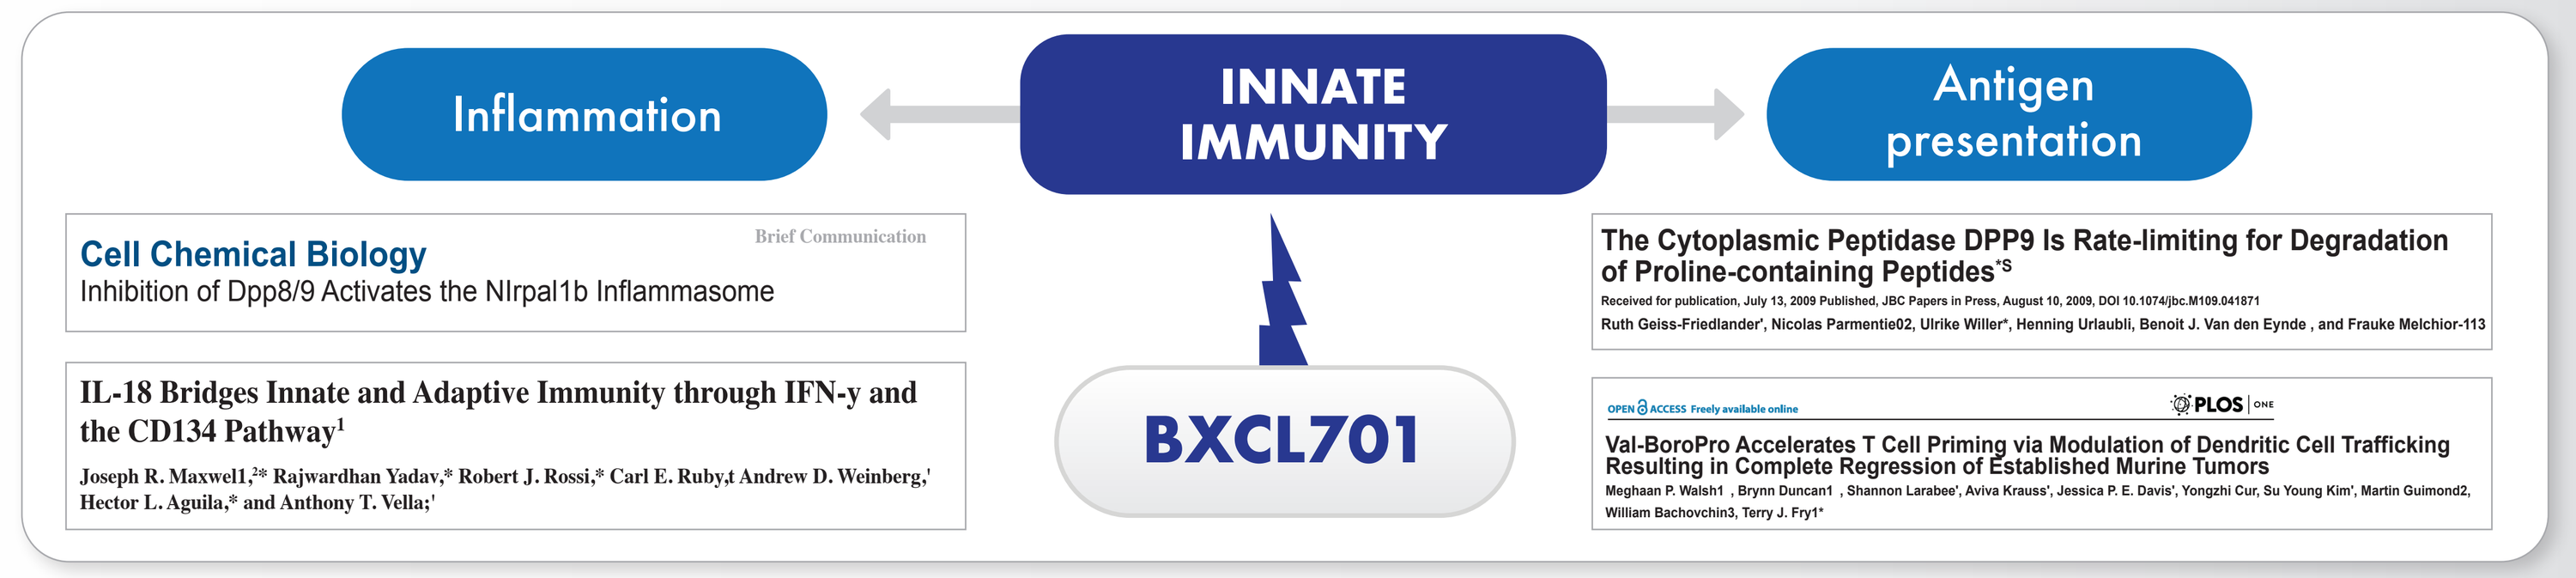
Inflammation
INNATE IMMUNITY
Antigen presentation
BXCL701
Cell Chemical Biology
Brief Communication
Inhibition of Dpp8/9 Activates the Nlrpa1b Inflammasome
IL-18 Bridges Innate and Adaptive Immunity through IFN-γ and the CD134 Pathway
The Cytoplasmic Peptidase DPP9 Is Rate-limiting for Degradation of Proline-containing Peptides
Val-BoroPro Accelerates T Cell Priming via Modulation of Dendritic Cell Trafficking Resulting in Complete Regression of Established Murine Tumors

MACROPHAGE PYROPTOSIS: THE CENTRAL COMPONENT OF BXCL701 ACTIVITY THROUGH DPP8/9 INHIBITION

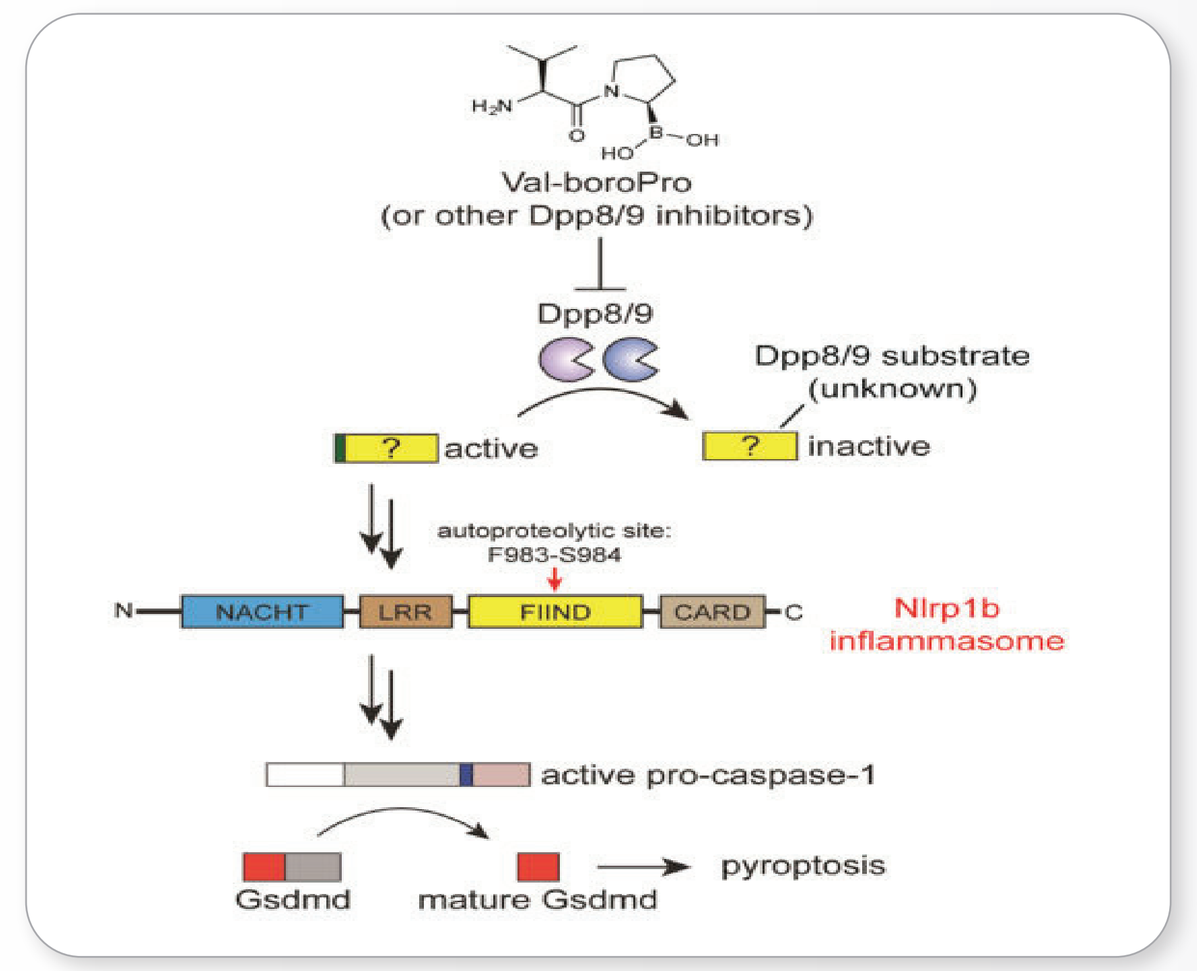
Val-boroPro
(or other Dpp8/9 inhibitors)
Dpp8/9
Dpp8/9 substrate (unknown)
active
inactive
autoproteolytic site: F983-S984
NACHT
LRR
FIIND
CARD
Nlrp1b inflammasome
active pro-caspase-1
pyroptosis
Gsdmd
mature Gsdmd

- Inhibition of DPP8/9 has been shown to activate the pro-inflammatory process of pyroptosis in macrophages
- Macrophage pyroptosis through the Nlrp1b pathway has been shown to drive the activation of caspase-1 and subsequent activation of pro-IL-18 and pro-IL-1β leading to the production of a host of cytokines and
- chemokines including IL-18 and IL-1β
 Mouse genetics further supports this process as the driver of BXCL701 activity via inhibition of DPP8/9

Okondo MC et al. Cell Chem Biol. 2018

PRECLINICAL DATA VALIDATES BXCL701 AND ANTI-PD-1 COMBINATION POTENTIAL

BXCL701/ANTI-PD-1 COMBINATION SHOWED:

- Synergistic antitumor effect on tumor volume in MC38 mouse model of colon adenocarcinoma
- Synergistic upregulation in the immunomodulatory parameters for pro-inflammatory cytokines*, chemokines+ and associated memory T-cells# by dose as single agent and in combination
- Synergistic increase in the cytotoxic NK cells and macrophages in the tumor with a decrease in the immunosuppressive T-regs

AACR 2017 Abstract 2629

*IL-2, IL-12p40, IL-6; +GM-CSF, G-CSF; #IL-15, IL-7

TRIAL SCHEMATIC AND KEY OBJECTIVES

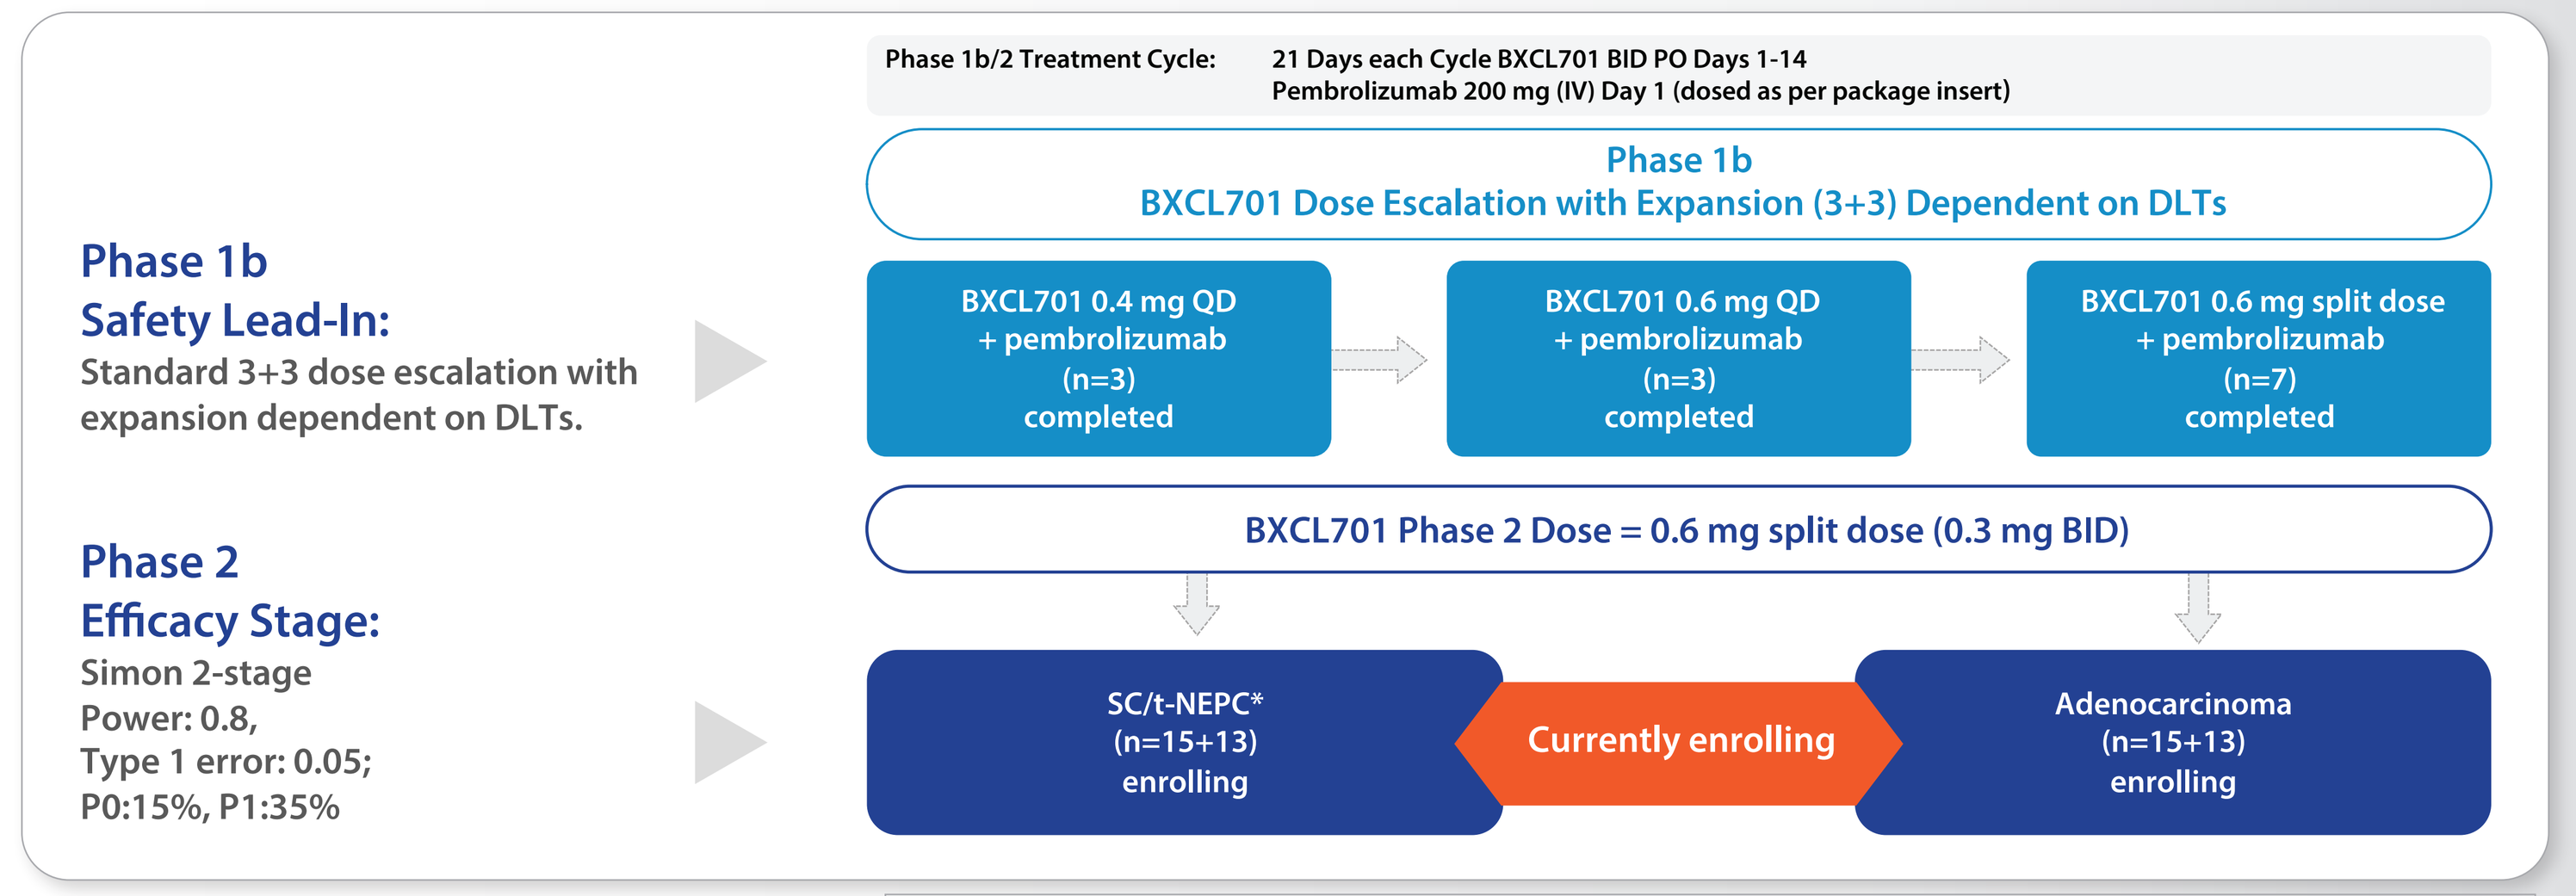
Phase 1b
Safety Lead-In:
Standard 3+3 dose escalation with expansion dependent on DLTs.
Phase 2
Efficacy Stage:
Simon 2-stage
Power: 0.8,
Type 1 error: 0.05;
P0:15%, P1:35%
Phase 1b/2 Treatment Cycle: 21 Days each Cycle BXCL701 BID PO Days 1-14
Pembrolizumab 200 mg (IV) Day 1 (dosed as per package insert)
Phase 1b
BXCL701 Dose Escalation with Expansion (3+3) Dependent on DLTs
BXCL701 0.4 mg QD + pembrolizumab (n=3) completed
BXCL701 0.6 mg QD + pembrolizumab (n=3) completed
BXCL701 0.6 mg split dose + pembrolizumab (n=7) completed
BXCL701 Phase 2 Dose = 0.6 mg split dose (0.3 mg BID)
SC/t-NEPC* (n=15+13) enrolling
Currently enrolling
Adenocarcinoma (n=15+13) enrolling

*Small-Cell/Treatment emergent neuroendocrine prostate cancer

Primary Objective for each group:
Composite Response Rate: Target >15%
Need >2 composite responses to proceed to Stage 2

Additional Objectives:
DoR, PFS, changes in circulating cytokines and correlation of outcome with baseline tumor characteristics

KEY INCLUSION AND EXCLUSION CRITERIA

KEY PHASE 1b INCLUSION CRITERIA

- Progression as defined by PCWG3 criteria
- At least 1 prior line of systemic therapy for locally advanced or metastatic prostate cancer
- Serum testosterone <50 ng/dL during Screening except for those with de novo small cell prostate cancer
- Eastern Cooperative Oncology Group (ECOG) performance status of 0-2

KEY PHASE 1b EXCLUSION CRITERIA

- More than 2 cytotoxic chemotherapy regimens for CRPC
- Prior treatment with an anti-PD-1, anti-PD-L1, anti-programmed death-ligand 2 (PD-L2) agent or with an agent directed to another co-inhibitory T-cell receptor
- Additional active malignancy that may confound the assessment of the study endpoints
- Brain metastases that are symptomatic and progressive on imaging
- Significant cardiovascular or pulmonary disease

See ClinicalTrials.gov Identifier: NCT03910660 for more details

PHASE 1b RESULTS

Baseline Characteristics		No. of Subjects: BXCL701 0.4 mg qd + pembrolizumab (N=3)	BXCL701 0.6 mg qd + pembrolizumab (N=3)	BXCL701 0.6 mg split dose + pembrolizumab (N=7)	All (N=13)
Age	Mean (range), years	70 (61-75)	71 (62-86)	73 (62-84)	72 (61-86)
ECOG PS 0/1/2 (n)	PS 0/1/2 (n)	2/1/0	2/1/0	1/6/0	5/8/0
Most Recent Histopathology	Adenocarcinoma	3	2	2	7
	SC/t-NEPC	-	1	5	6
Prior Systemic Therapies	Any ADT therapy	2	3	5	10
	1st generation ADT therapy	1	2	3	6
	2nd generation ASI	2	2	5	9
	Chemotherapy	3	1	7	11
	Radiotherapy	2	3	6	11

ADT= Androgen deprivation therapy; ASI = Androgen signaling inhibitor | 30-Sep-2020

EXPOSURE DURATION AND SUBJECT DISPOSITION

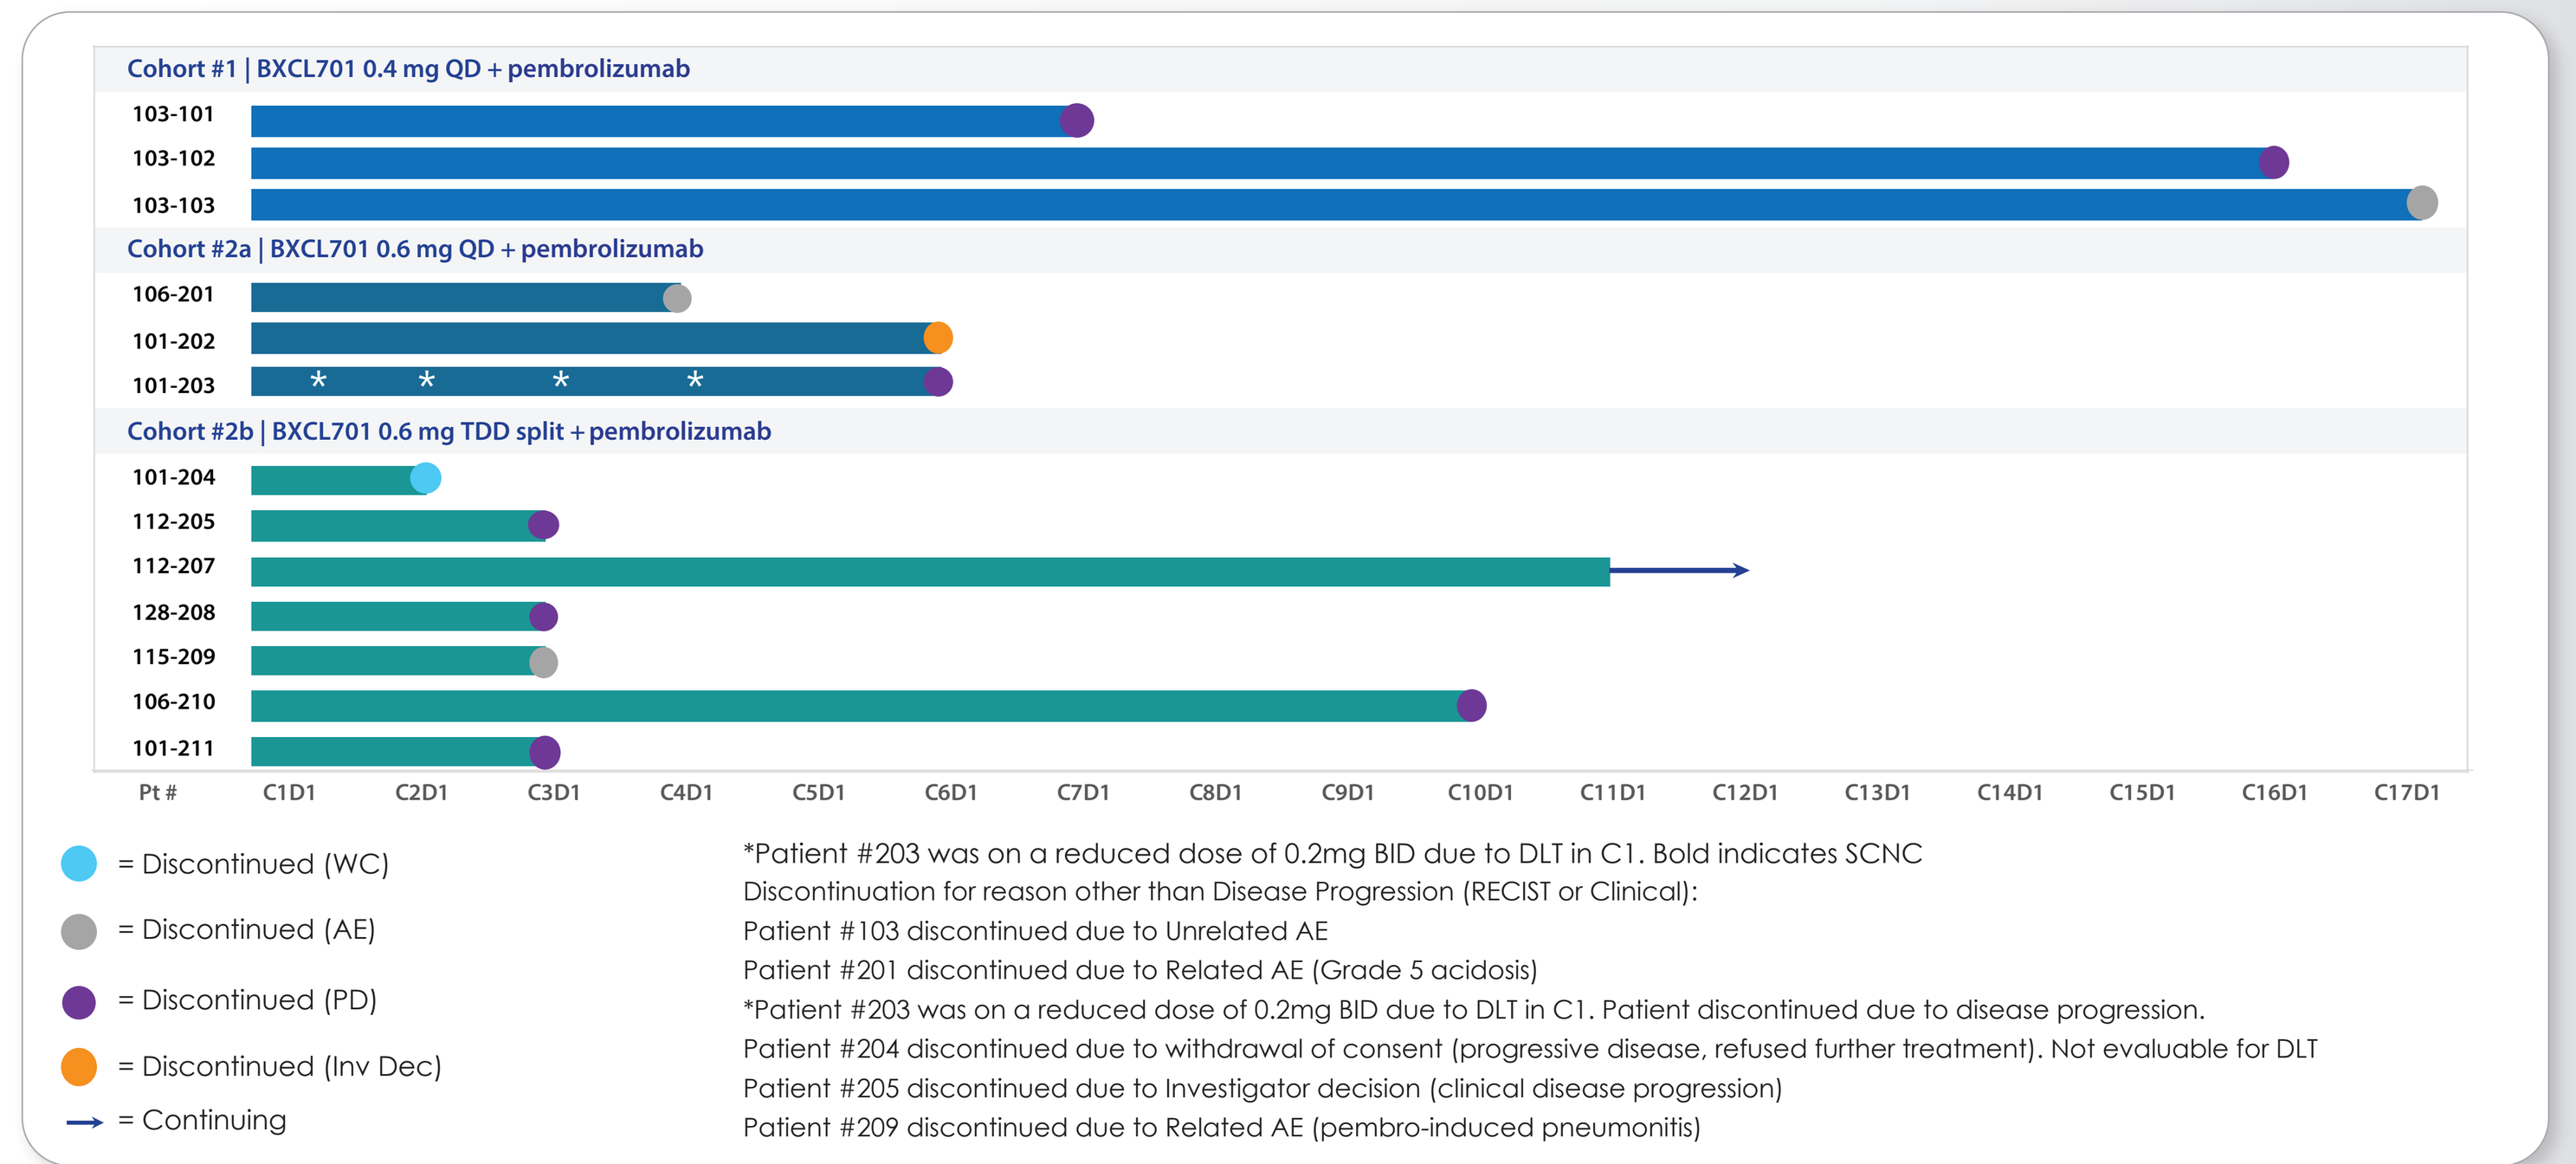
Cohort #1 | BXCL701 0.4 mg QD + pembrolizumab
103-101
103-102
103-103
Cohort #2a | BXCL701 0.6 mg QD + pembrolizumab
106-201
101-202
101-203
Cohort #2b | BXCL701 0.6 mg TDD split + pembrolizumab
101-204
112-205
112-207
128-208
115-209
106-210
101-211
Pt #
C1D1 C2D1 C3D1 C4D1 C5D1 C6D1 C7D1 C8D1 C9D1 C10D1 C11D1 C12D1 C13D1 C14D1 C15D1 C16D1 C17D1
= Discontinued (WC)
= Discontinued (AE)
= Discontinued (PD)
= Discontinued (Inv Dec)
= Continuing

*Patient #203 was on a reduced dose of 0.2mg BID due to DLT in C1. Bold indicates SCNC
Discontinuation for reason other than Disease Progression (RECIST or Clinical):
Patient #103 discontinued due to Unrelated AE
Patient #201 discontinued due to Related AE (Grade 5 acidosis)
*Patient #203 was on a reduced dose of 0.2mg BID due to DLT in C1. Patient discontinued due to disease progression.
Patient #204 discontinued due to withdrawal of consent (progressive disease, refused further treatment). Not evaluable for DLT
Patient #205 discontinued due to Investigator decision (clinical disease progression)
Patient #209 discontinued due to Related AE (pembro-induced pneumonitis)

BXCL701 ON-TARGET ADVERSE EVENTS CONSISTENT WITH CYTOKINE ACTIVATION SEEN AT THE HIGHEST DOSES

Preferred Term	BXCL701 0.4 mg qd + pembrolizumab N=3				BXCL701 0.6 mg qd + pembrolizumab N=3				BXCL701 0.6 mg Split dose + pembrolizumab N=7			
	(Grade) N											
BXCL701 Related Events#	(Any)	(1)	(2)	(3)	(Any)	(1)	(2)	(3)	(Any)	(1)	(2)	(3)
Hypotension*	-	-	-	-	3	2	1	-	1	1	-	-
Dizziness	-	-	-	-	2	2	-	-	-	-	-	-
Headache	-	-	-	-	1	1	-	-	-	-	-	-
Syncope	-	-	-	-	1	-	-	1	-	-	-	-
Dyspnea	-	-	-	-	1	1	-	-	1	-	1	-
Chills	-	-	-	-	-	-	-	-	1	-	1	-
Edema	-	-	-	-	2	1	1	-	-	-	-	-
Fatigue	-	-	-	-	1	-	1	-	3	2	-	1†
Myalgia	-	-	-	-	1	1	-	-	1	1	-	-

Relatedness as assessed by investigator, *Includes orthostatic hypotension, † Fatigue present at baseline

Other BXCL701 Related Adverse Events Grade ≥3 (1 acidosis) or Low Grade in ≥2 patients (arthralgia, hematologic, decrease appetite)

EFFICACY RESULTS

Responses	BXCL701 0.4 mg qd + pembrolizumab N=3	BXCL701 0.6 mg qd + pembrolizumab N=3	BXCL701 0.6 mg split dose + pembrolizumab N=7
RECIST 1.1			
SD	2	1	1
Non-CR/Non-PD	1	-	1
PD	-	1	2
NE	-	1*	3*
PSA Evaluable	3	2	5
PSA Response†	0	0	1

† ≥50% decline by week 12
* Early death or early DC without follow-up disease assessment

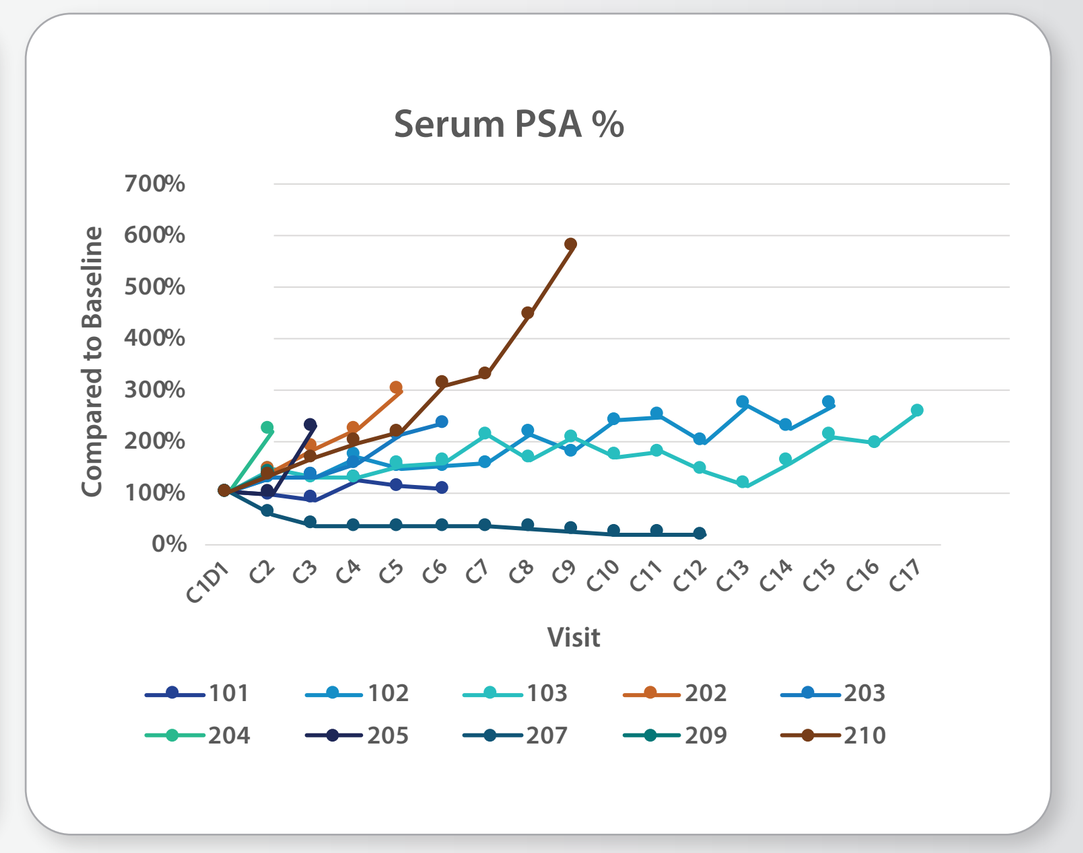
Serum PSA %
Compared to Baseline
700%
600%
500%
400%
300%
200%
100%
0%
C1D1 C2 C3 C4 C5 C6 C7 C8 C9 C10 C11 C12 C13 C14 C15 C16 C17
Visit
101
102
103
202
203
204
205
207
209
210

CONFIRMED PSA RESPONDER

- 73 years, diagnosed 2006 with Adenocarcinoma; multiple lymph nodes and bone lesions; Baseline PSA 163.1ng/mL
- Duration of BXCL701+ pembrolizumab: 11 cycles
- Prior Systemic Therapy included: Bicalutamide; Abiraterone + Prednisone; Enzalutamide; Sipuleucel Docetaxel/Cabazitaxel
- Confirmed PSA Response:
 Time to PSA response: 6 weeks (60.4 ng/mL - 63% reduction), ongoing at 30 weeks (25.9 ng/mL - 84% reduction) and ongoing
- RECIST 1.1 Status: Minor Response
 - Target Lesions: minor response ongoing (22% reduction 51 mm > 40 mm)
 - Non-Target Lesions: Disappearance of 2/4 lymph nodes and non-PD/non-CR bone disease

CYTOKINE DATA FROM PHASE IB SAFETY LEAD IN COHORTS

IL-18 DATA

Mechanistic relevance of IL-18: IL-18 is produced mainly by macrophages, induces NK cell activation and IFN-γ in T cells and acts as a bridge between innate and adaptive immunity, favouring anti-tumor immunity

RESULTS

- IL-18 levels generally increased post dose in all the dose cohorts (0.4 mg qd, 0.6 mg qd and 0.6 mg split dose in combination with pembrolizumab)
- 0.6 mg split dose + pembrolizumab showed consistent increases in IL-18 on day 14 post dose (8-24 fold)
- 106-201 (0.6 mg QD + pembrolizumab) showed maximum IL-18 levels (45 fold)

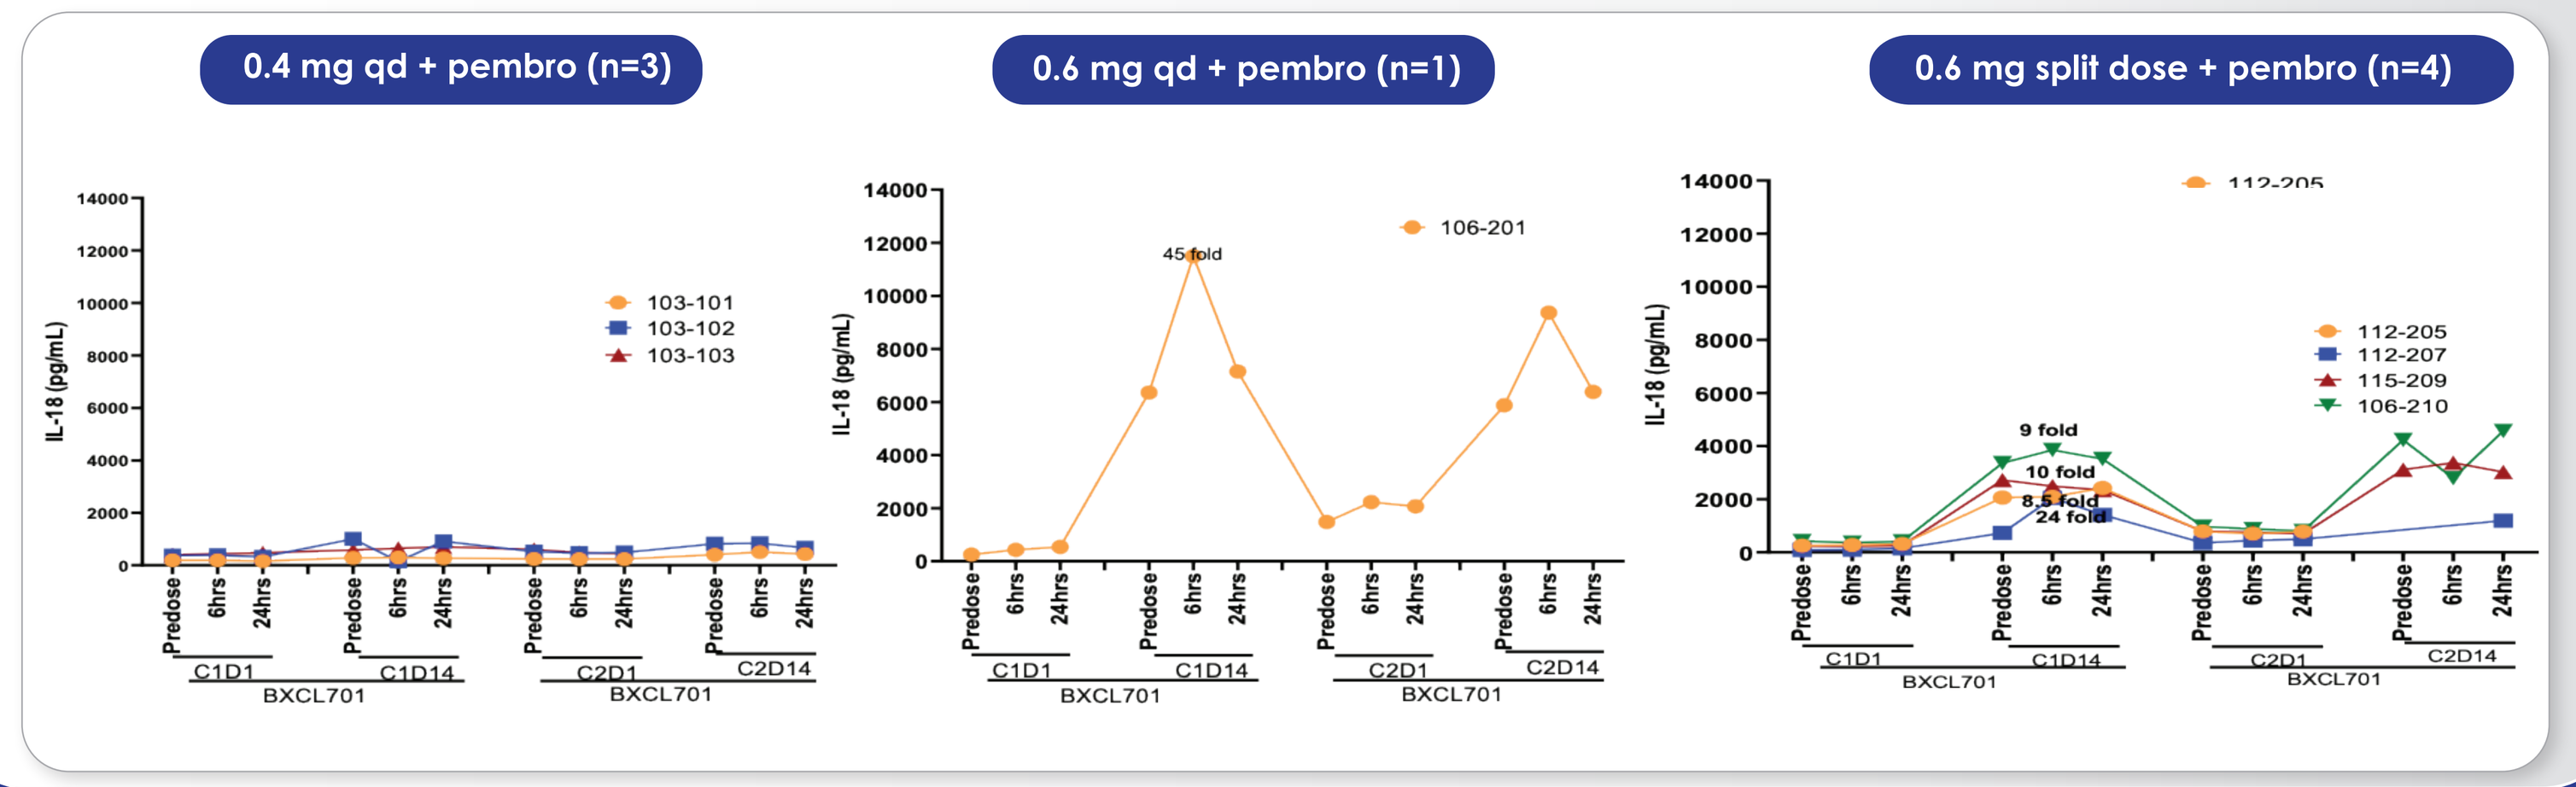
0.4 mg qd + pembro (n=3)
0.6 mg qd + pembro (n=1)
0.6 mg split dose + pembro (n=4)
IL-18 (pg/mL)
103-101
103-102
103-103
106-201
45 fold
112-205
112-207
115-209
106-210
9 fold
10 fold
8.5 fold
24 fold
Predose 6hrs 24hrs
C1D1 C1D14 C2D1 C2D14
BXCL701

IFN-γ CHANGES POST DOSE

IFN-γ produced by activated T cells and NK cells; high levels are associated with effective host defense and antitumor immunity

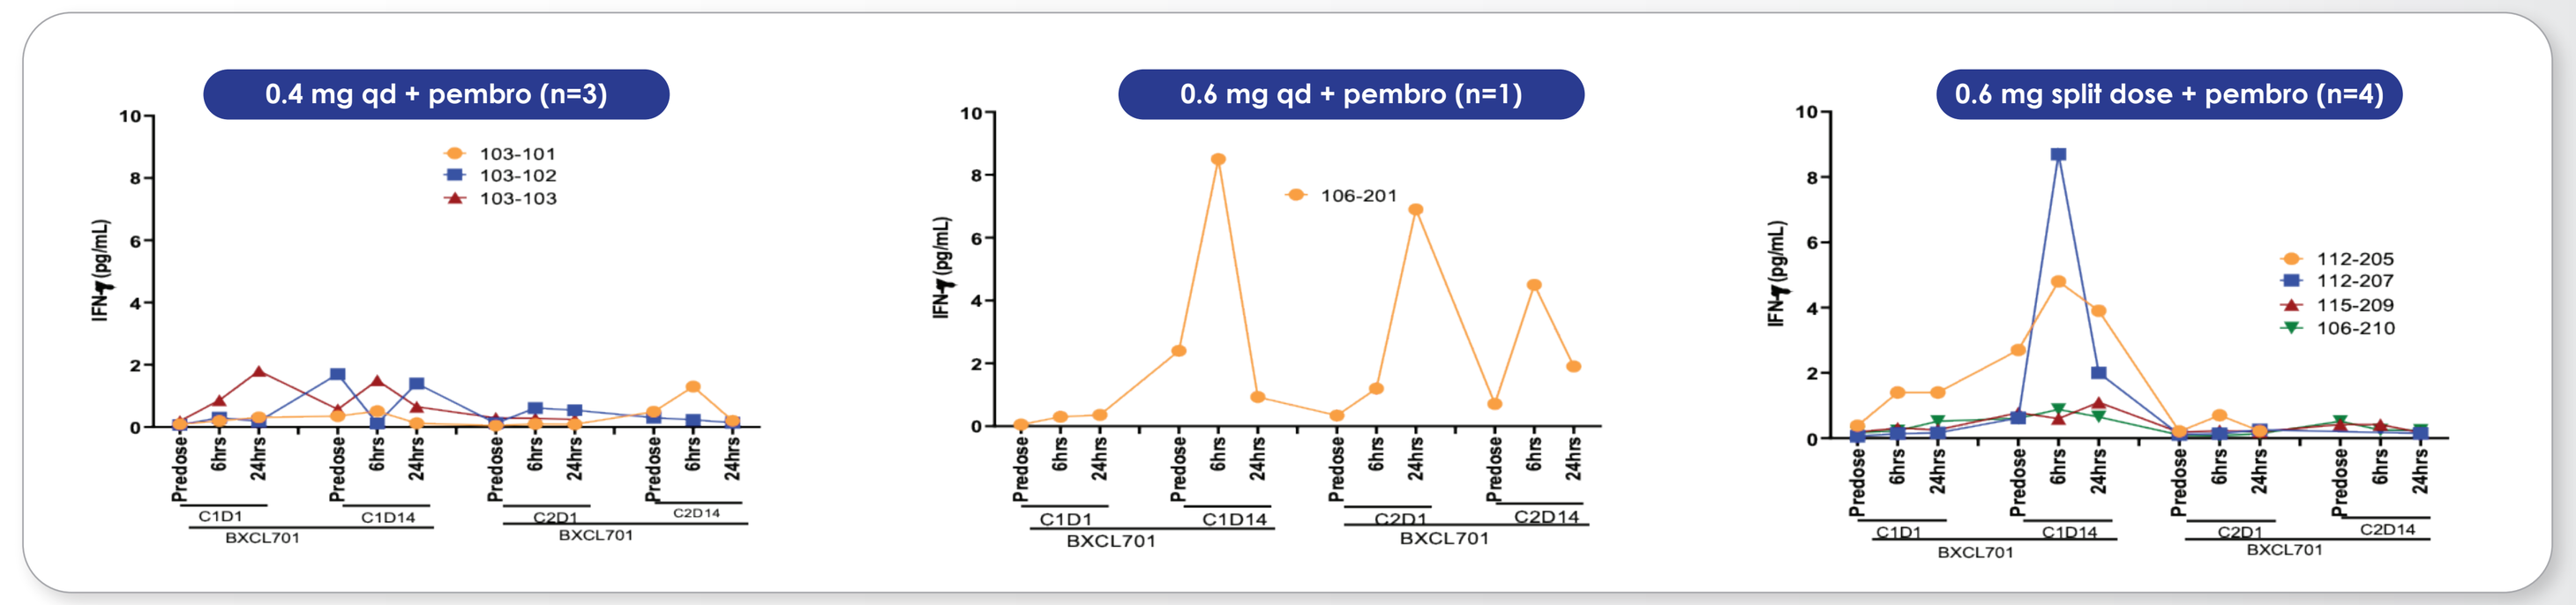
0.4 mg qd + pembro (n=3)
0.6 mg qd + pembro (n=1)
0.6 mg split dose + pembro (n=4)
IFN-γ (pg/mL)
103-101
103-102
103-103
106-201
112-205
112-207
115-209
106-210
Predose 6hrs 24hrs
C1D1 C1D14 C2D1 C2D14
BXCL701

- Time dependent increase in IFN-γ levels observed in 0.6 mg qd and split dose cohorts.

IL-1β CHANGES POST DOSE

IL-1β is a proinflammatory cytokine produced by innate immune cells. IL-1β is associated with safety and is pro-tumorigenic

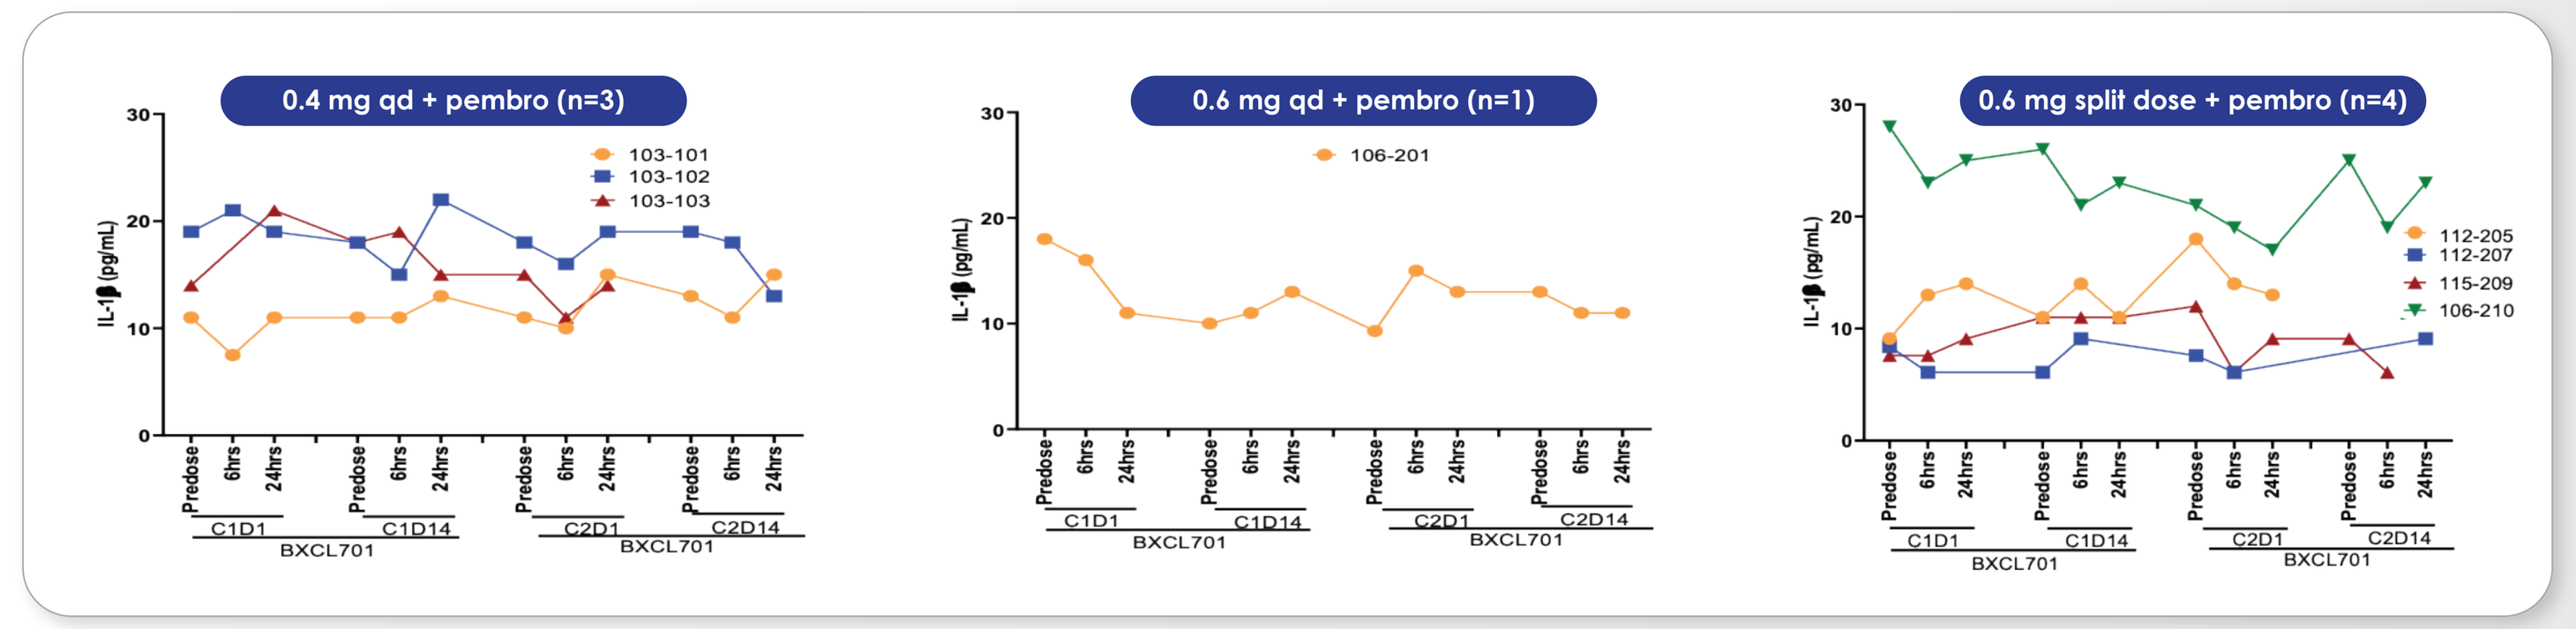
0.4 mg qd + pembro (n=3)
0.6 mg qd + pembro (n=1)
0.6 mg split dose + pembro (n=4)
IL-1β (pg/mL)
103-101
103-102
103-103
106-201
112-205
112-207
115-209
106-210
Predose 6hrs 24hrs
C1D1 C1D14 C2D1 C2D14
BXCL701

- IL-1β levels did not demonstrates a trend in 0.4 mg QD + pembrolizumab dose group neither in 0.6 mg QD and split dose cohorts during dosing

IL-6 (0.6 mg SPLIT DOSE + PEMBROLIZUMAB)

- IL-6 is a pleiotropic cytokine. IL-6 helps tumor cell proliferation. Increased IL-6 associated with cytokine release syndrome
- In the majority of patients, any increase in IL-6 quickly returned to baseline

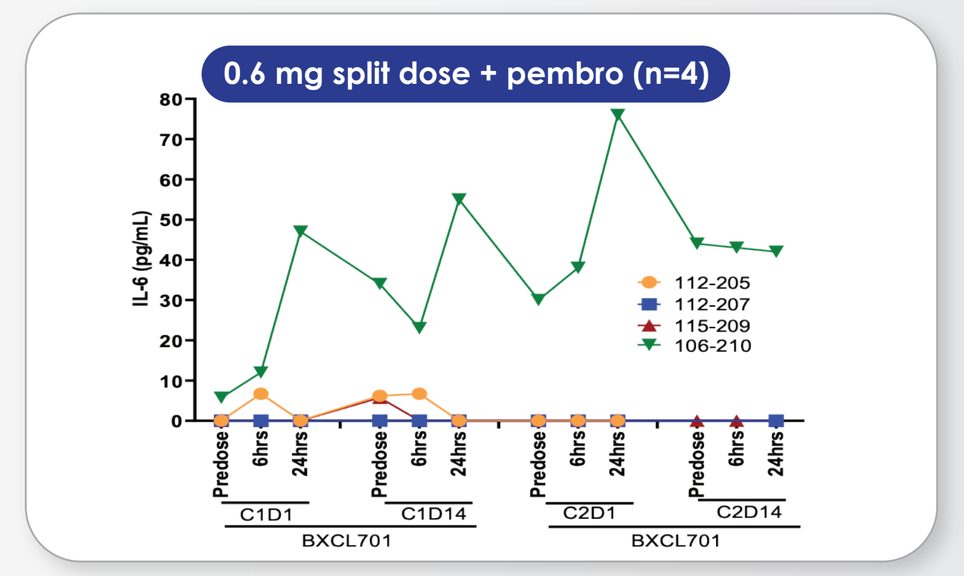
0.6 mg split dose + pembro (n=4)
IL-6 (pg/mL)
112-205
112-207
115-209
106-210
Predose 6hrs 24hrs
C1D1 C1D14 C2D1 C2D14
BXCL701

IL-8 (0.6 mg SPLIT DOSE + PEMBROLIZUMAB)

- IL-8, neutrophil chemotactic factor, induces chemotaxis in neutrophils and granulocytes. Increased IL-8 associated with safety adverse events
- Post-treatment IL-8 levels remained similar to baseline levels

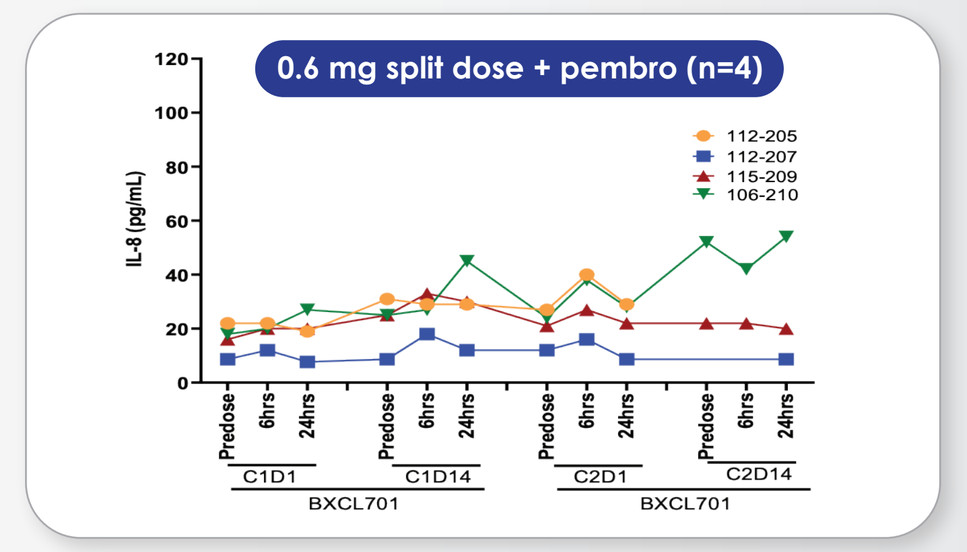
0.6 mg split dose + pembro (n=4)
IL-8 (pg/mL)
112-205
112-207
115-209
106-210
Predose 6hrs 24hrs
C1D1 C1D14 C2D1 C2D14
BXCL701

CONCLUSION

- Results from the Phase 1b safety assessment of BXCL701 support the use of a split dose totaling 0.6 mg per day as the recommended dose when used in combination with pembrolizumab
- On-target adverse events consistent with cytokine activation were seen at the highest daily dose tested
 - Splitting the daily dose was associated with improved tolerability as evidenced by no reported DLTs and lower rates of other adverse events of interest such as hypotension and peripheral edema
 - Consistent dose and time dependent increases in IL-18 and IFN-γ levels observed in 0.6 mg split dose, maximal changes noted after 14 days of dosing. Minimal and short-duration changes noted in cytokines often associated with AEs
 - Anti-tumor activity seen in 1 patient with a PSA response and four patients with stable disease per RECIST 1.1
- The Phase 2 portion of this study is currently enrolling patients with SC/t-NEPC or adenocarcinoma and will assess the anti-tumor activity of the combination in a setting where checkpoint inhibitor monotherapies have demonstrated limited clinical benefit

THANK YOU

BioXcel Therapeutics would like to thank all patients, their families, and caregivers who make this study possible. BioXcel would also like to thank the participating investigators and their staff for their support on this study and their dedication to their patients, despite the additional challenges as a circumstance of the COVID-19 pandemic.

SITC 2020—Society for Immunotherapy of Cancer 35th Anniversary Annual Meeting