Dipeptidyl Peptidase Inhibitor BXCL701 activates Innate Immunity followed by Adaptive Immunity on a Molecular and Cellular Level in a Mouse Model of Pancreatic Cancer

Veena Agarwal, John MacDougall, Shubhendu Trivedi, Dimple Bhatia, Zeenia Jagga, Hemant Banga, Diane Healey, Sreenivas Adurthi, Vincent O'Neill
BioXcel Therapeutics, 555 Long Wharf Drive, New Haven, CT, 06511

Abstract No #4189

BACKGROUND

- The oral DPP8/9 inhibitor BXCL701 in triple combination with aPD-1 and pegylated IL-2 (NKTR-214, bempegaldesleukin) has demonstrated complete regression in animal model of pancreatic cancer [ASCO 2018].
- BXCL701 is known to inhibit regulatory proteases DPP8/9, consequently activating Nlrp1b inflammasome, which in turn activates pro-caspase-1 to mediate pyroptosis in mouse macrophages [Okondo et al, 2018]. Caspase-1 is involved in cleavage of pro-IL-1β and pro-IL-18 to their active forms, IL-1β and IL-18 respectively [Bergsbaken et al. 2009].
- In the present study, the mechanism of action of BXCL701 has been elucidated in a sub-chronic pharmacokinetic / pharmacodynamic study at molecular and cellular level.
- BXCL701-treatment upregulates IL-18 markedly but not IL-1β. Other major upregulated cytokines were G-CSF, MIG, Eotaxin, MCP-1, IL-5, KC, IL-6 and IFN-γ.
- BXCL701 enhances T cells and NK cells infiltration along with expression of MHC genes within the tumor microenvironment.
- At the molecular level, innate and adaptive immune response genes, T-cell receptor genes and MHC genes were upregulated by BXCL701-treatment. Furthermore, genes associated with T cell and NK cell mediated apoptosis and cytolysis were upregulated and suggests enhanced cell death within tumor. On the other hand, downregulated gene clusters belonged to functional categories like cell cycle, DNA repair, several genes associated with cancer progression and extracellular matrix (ECM) modification.

EARLIER DATA DEMONSTRATED BXCL701 WITH PD-1 ANTAGONIST AND NKTR-214 GENERATED COMPLETE AND DURABLE RESPONSES

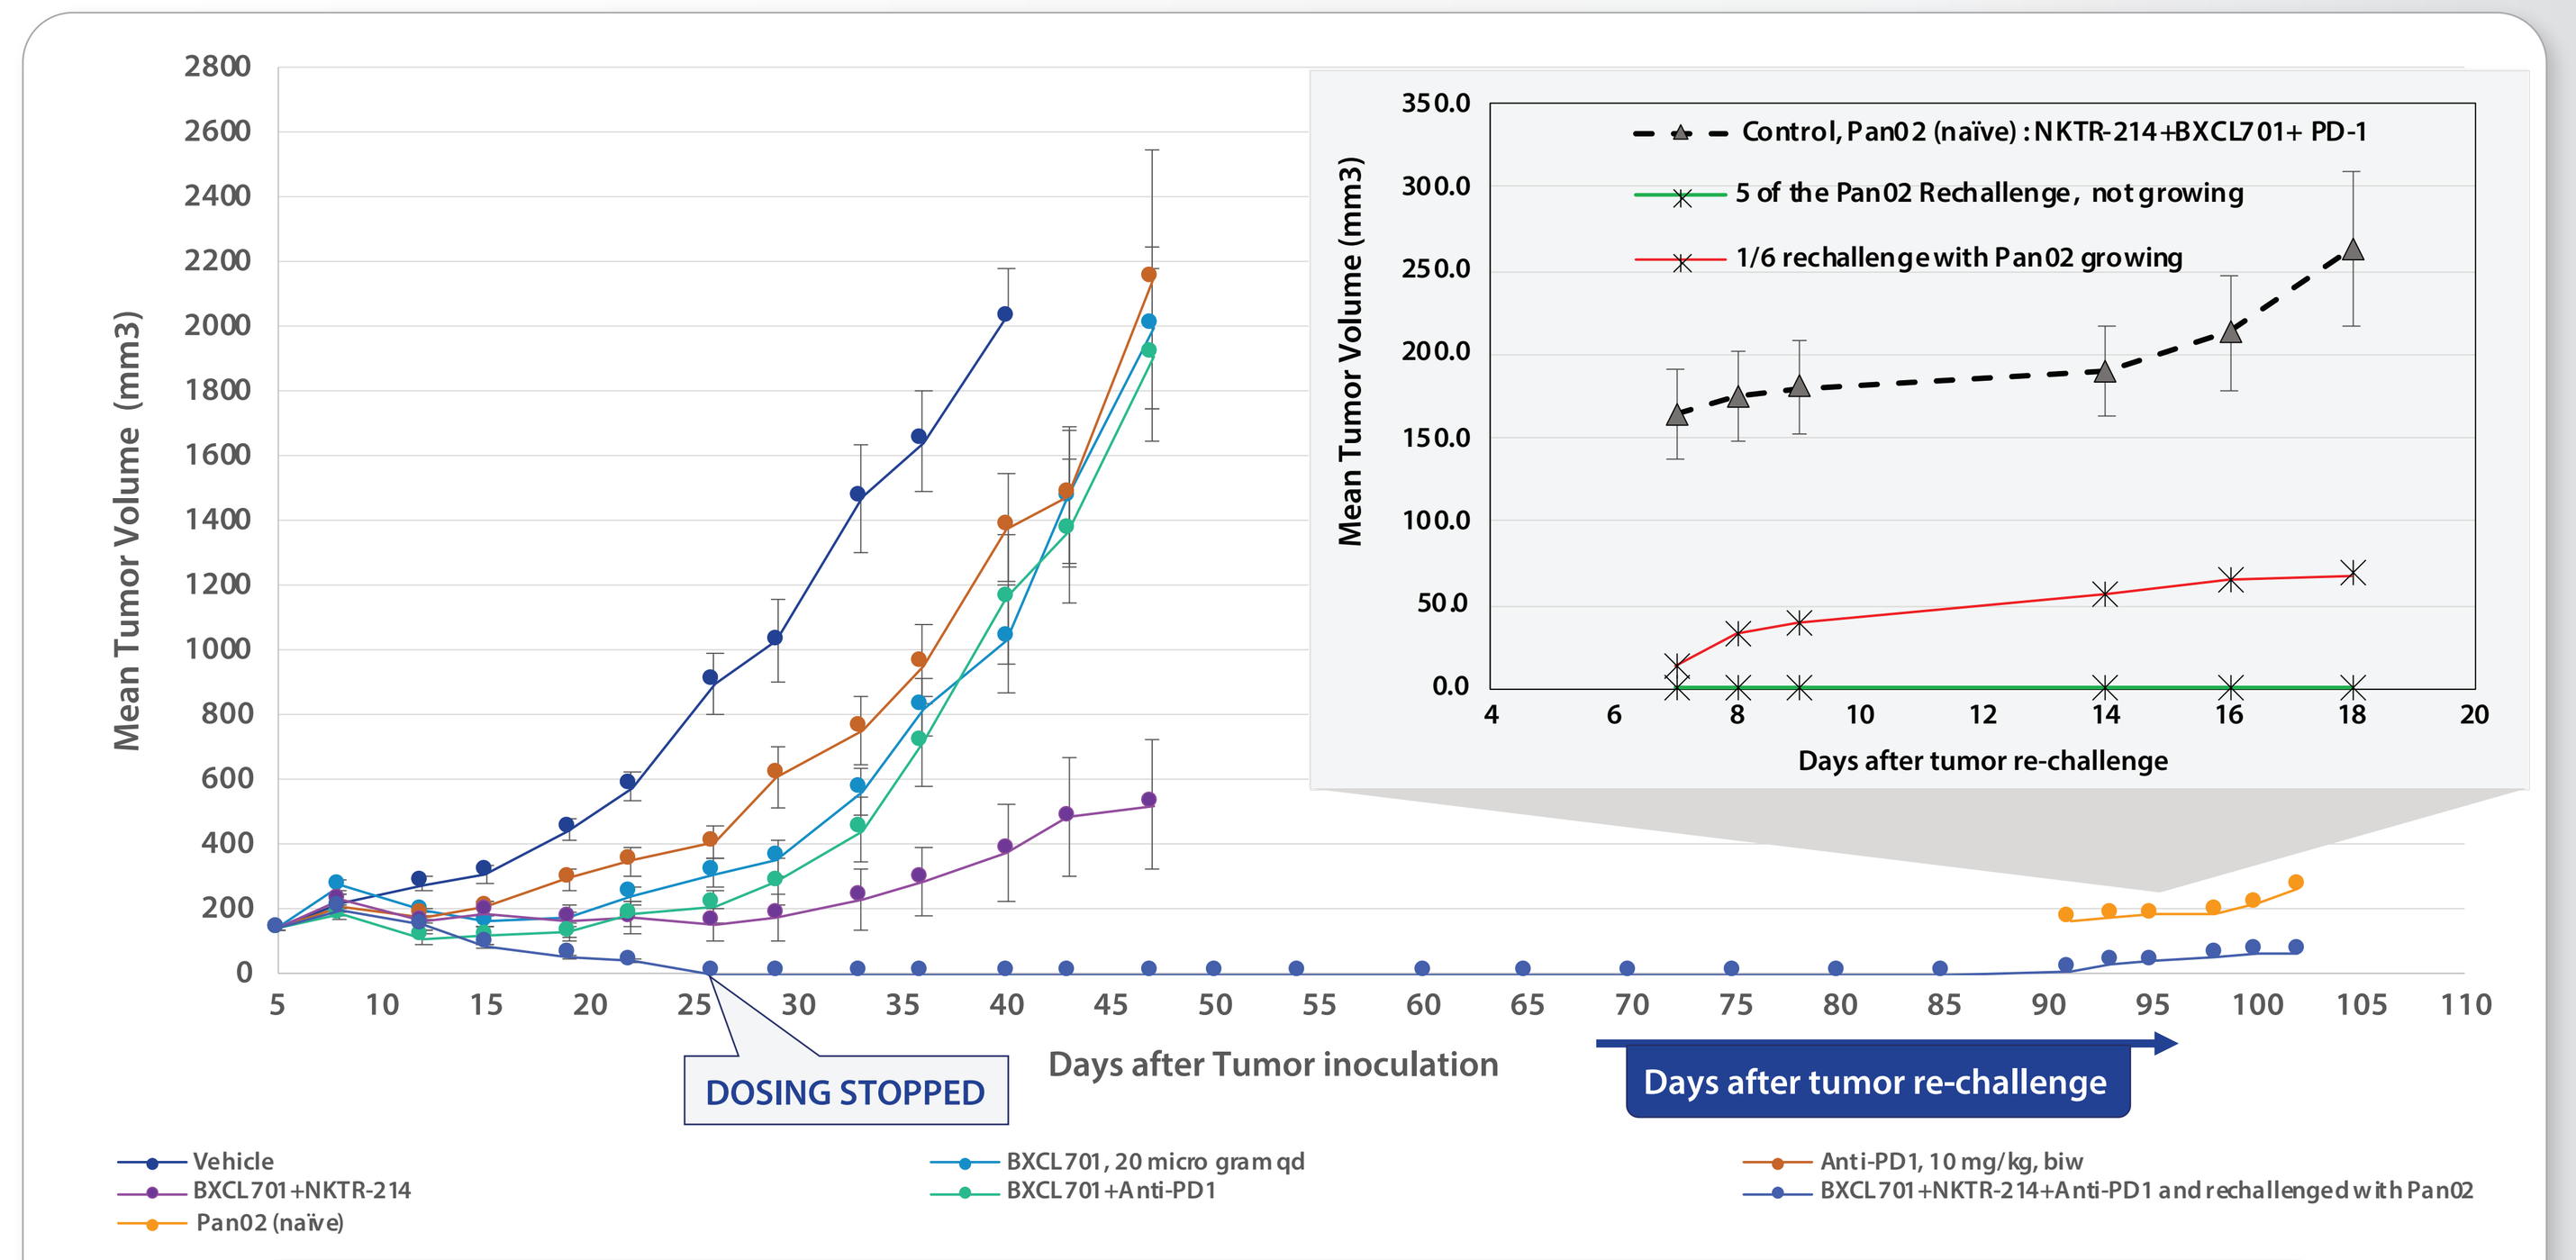
Mean Tumor Volume (mm3)
Days after Tumor inoculation
DOSING STOPPED
Days after tumor re-challenge
Control, Pan02 (naïve) : NKTR-214+BXCL701+ PD-1
5 of the Pan02 Rechallenge, not growing
1/6 rechallenge with Pan02 growing
Days after tumor re-challenge
Vehicle
BXCL701+NKTR-214
Pan02 (naive)
BXCL701, 20 micro gram qd
BXCL701+Anti-PD1
Anti-PD1, 10 mg/kg, biw
BXCL701+NKTR-214+Anti-PD1 and rechallenged with Pan02

Combo with anti-PD1 and NKTR-214 stimulates immune system "curing" mice and making them resistant to new tumors (Rastelli et all. 2018)

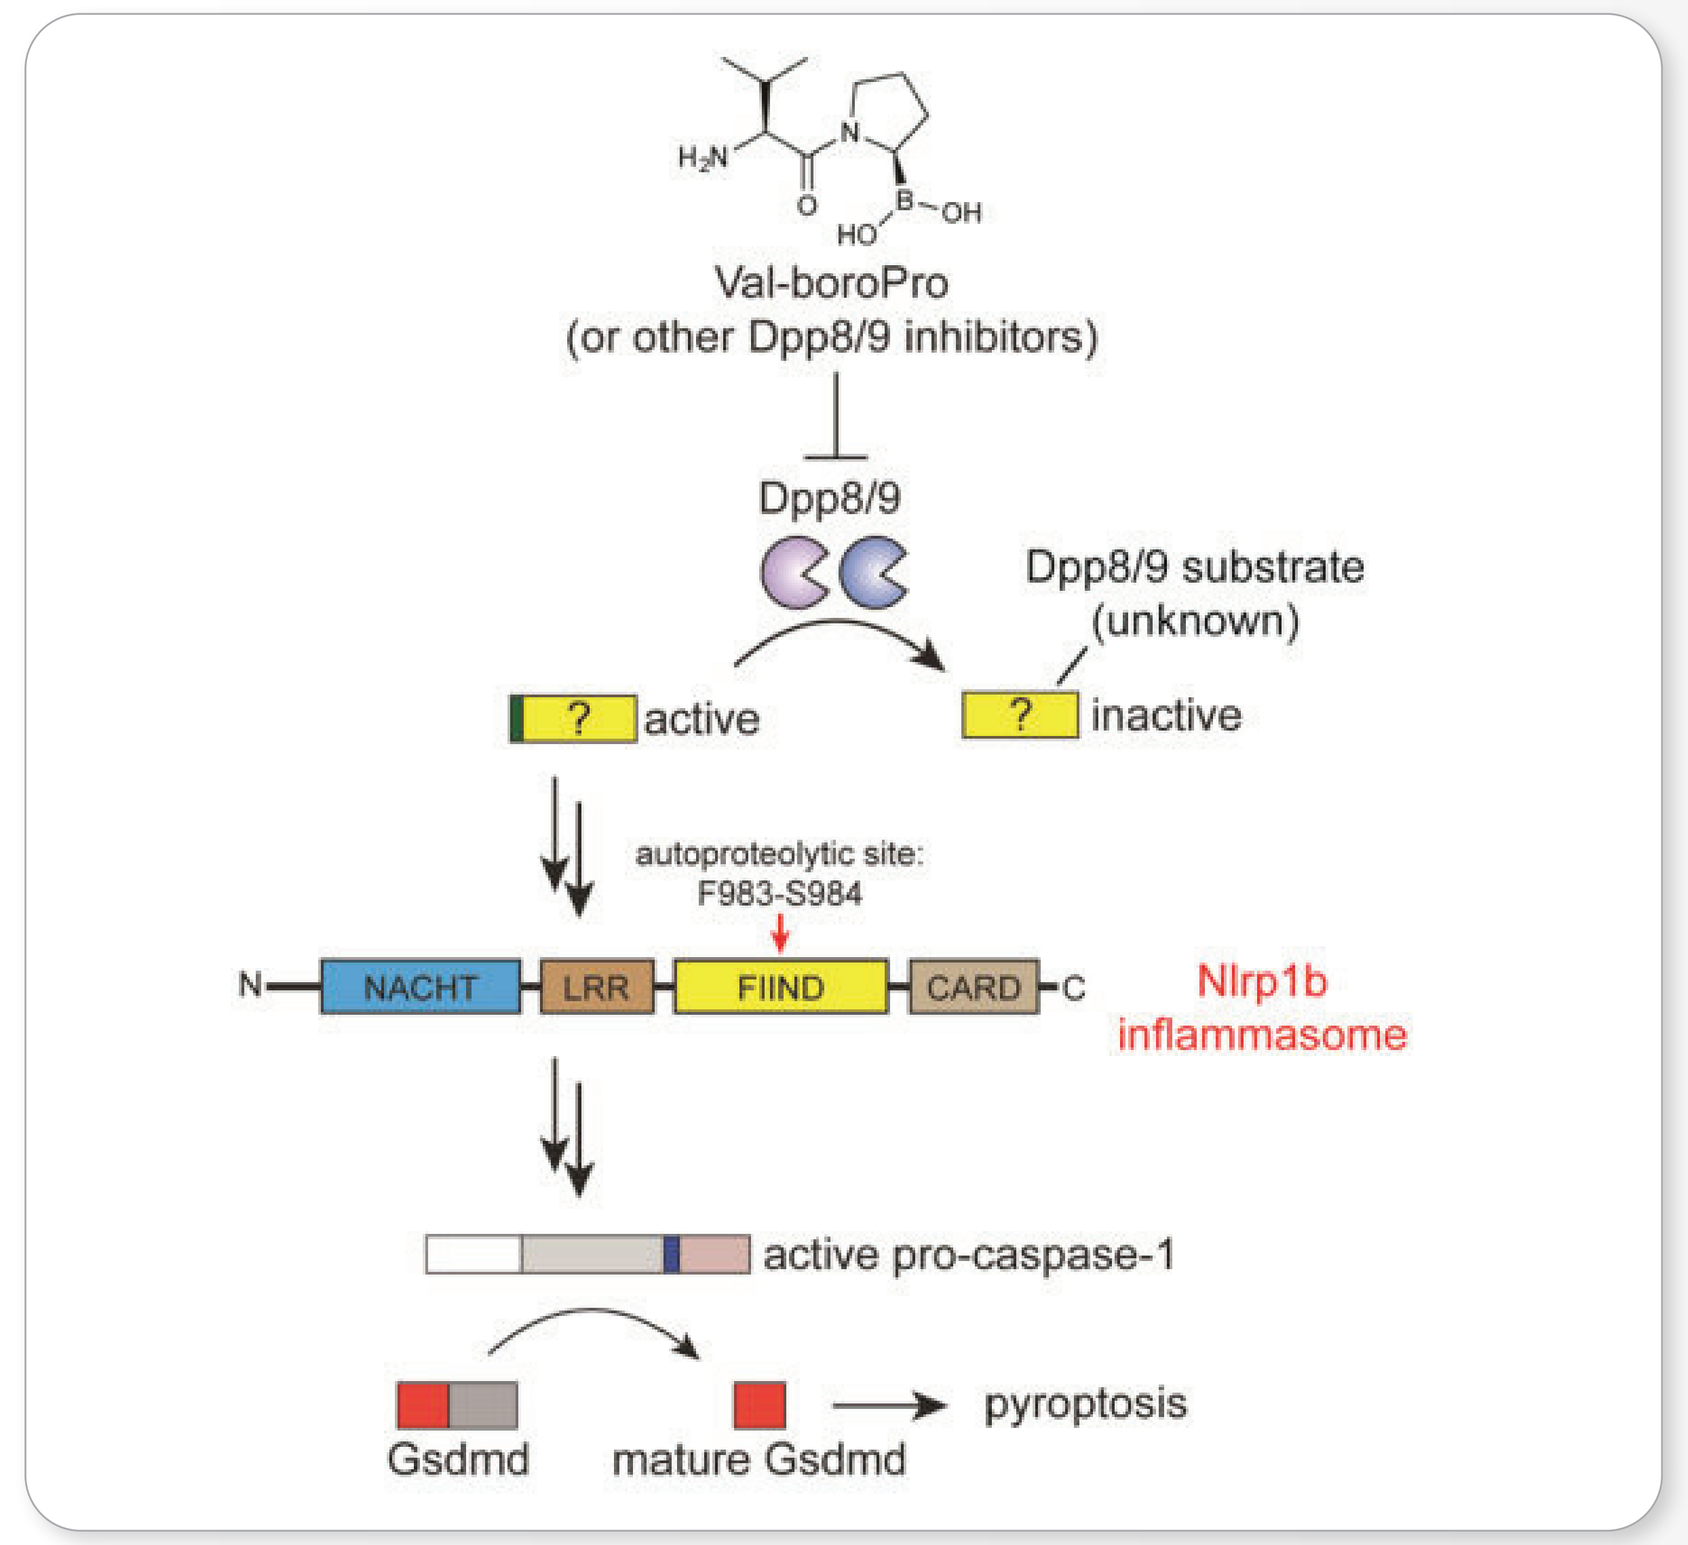
Val-boroPro
(or other Dpp8/9 inhibitors)
Dpp8/9
Dpp8/9 substrate (unknown)
? active
? inactive
autoproteolytic site: F983-S984
N NACHT LRR FIIND CARD C
Nlrp1b inflammasome
active pro-caspase-1
Gsdmd
mature Gsdmd
pyroptosis

DPP8/9 inhibition stimulates the innate immune system.

DPP8/9 inhibitors induce an inflammatory form of cell death called pyroptosis in monocytes and macrophages by activating the inflammosome sensor protein Nlrp1b which in turn activates pro-caspase -1 to mediate pyroptosis [Okondo et al. 2018].

METHODOLOGY - SUBCHRONIC PK/PD STUDY

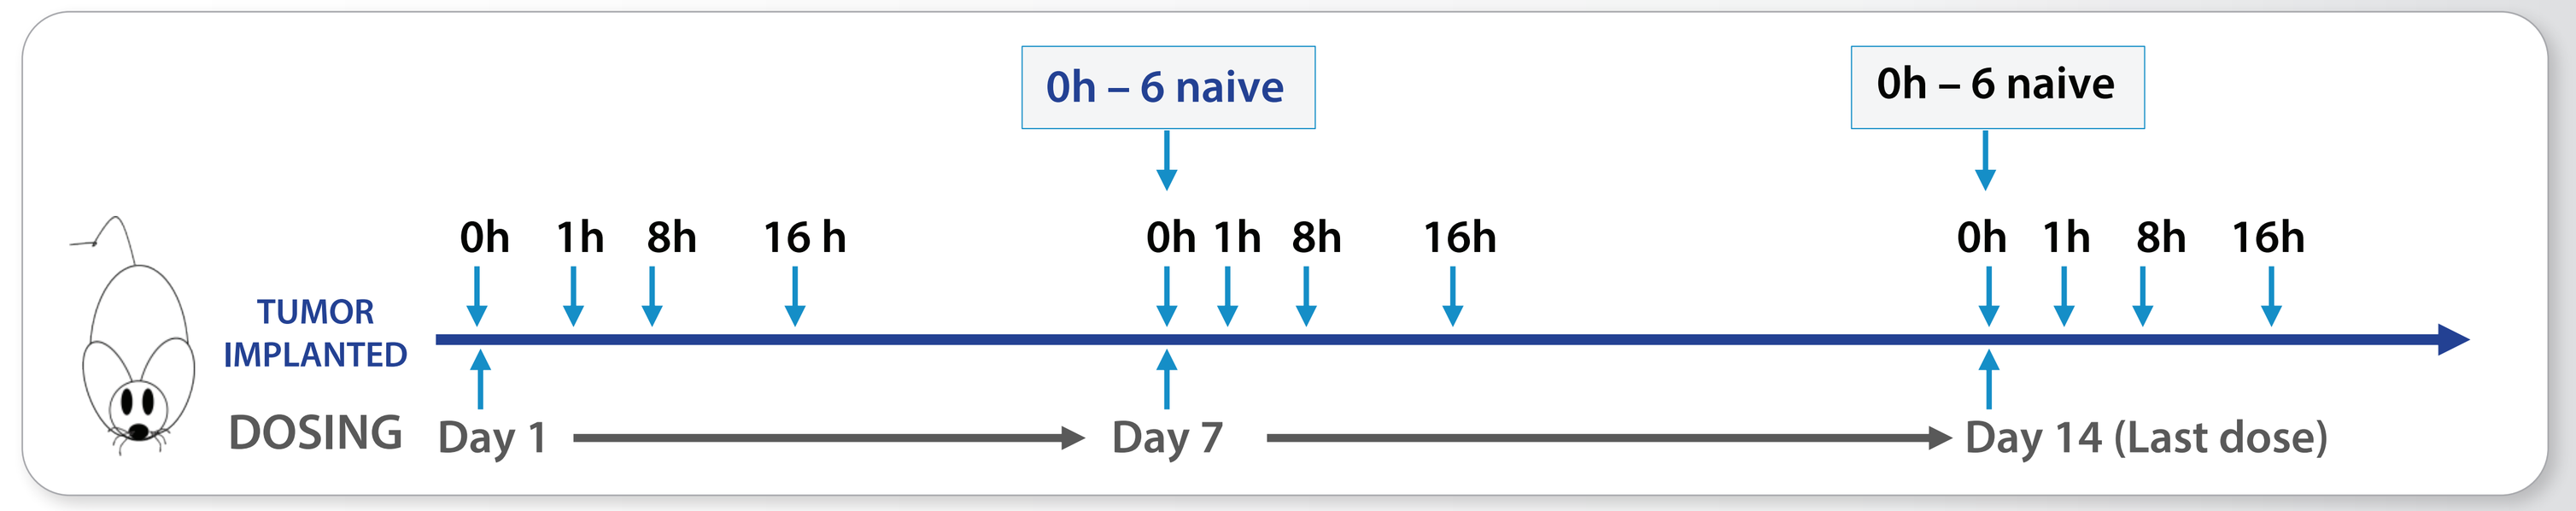
0h – 6 naive
0h – 6 naive
0h 1h 8h 16 h
0h 1h 8h 16h
0h 1h 8h 16h
TUMOR IMPLANTED
DOSING
Day 1
Day 7
Day 14 (Last dose)

Tumor model: : Pan02

Treatments: Vehicle and BXCL701 (20 µg, qd) were dosed for 14 days in mice bearing established (~200 mm3) tumors.

Tumor implant: Each mouse was inoculated subcutaneously at the front right flank region with an appropriate inoculum of cancer cells (approximately 3 x 10^6) in 0.1 ml of PBS for tumor development. The date of tumor cell inoculation is denoted as day 0. Approximately 20 days following tumor implant, mice with a mean tumor volume of ~ 200 mm3 were randomized and BXCL701 was administered.

Protocol: Cohorts of 6 animals were harvested at 0, 1, 8 and 16h after dosing at day 1, 7 and 14. Tumor and plasma samples were evaluated for cytokines by Luminex and ELISA, immunophenotypes by IHC, gene expression analyses by RNASeq, and PK. Tumor size and body weights were measured twice weekly.

CYTOKINE/CHEMOKINE UPREGULATION BY BXCL701 IN PANCREATIC CANCER MOUSE MODEL (PAN02)

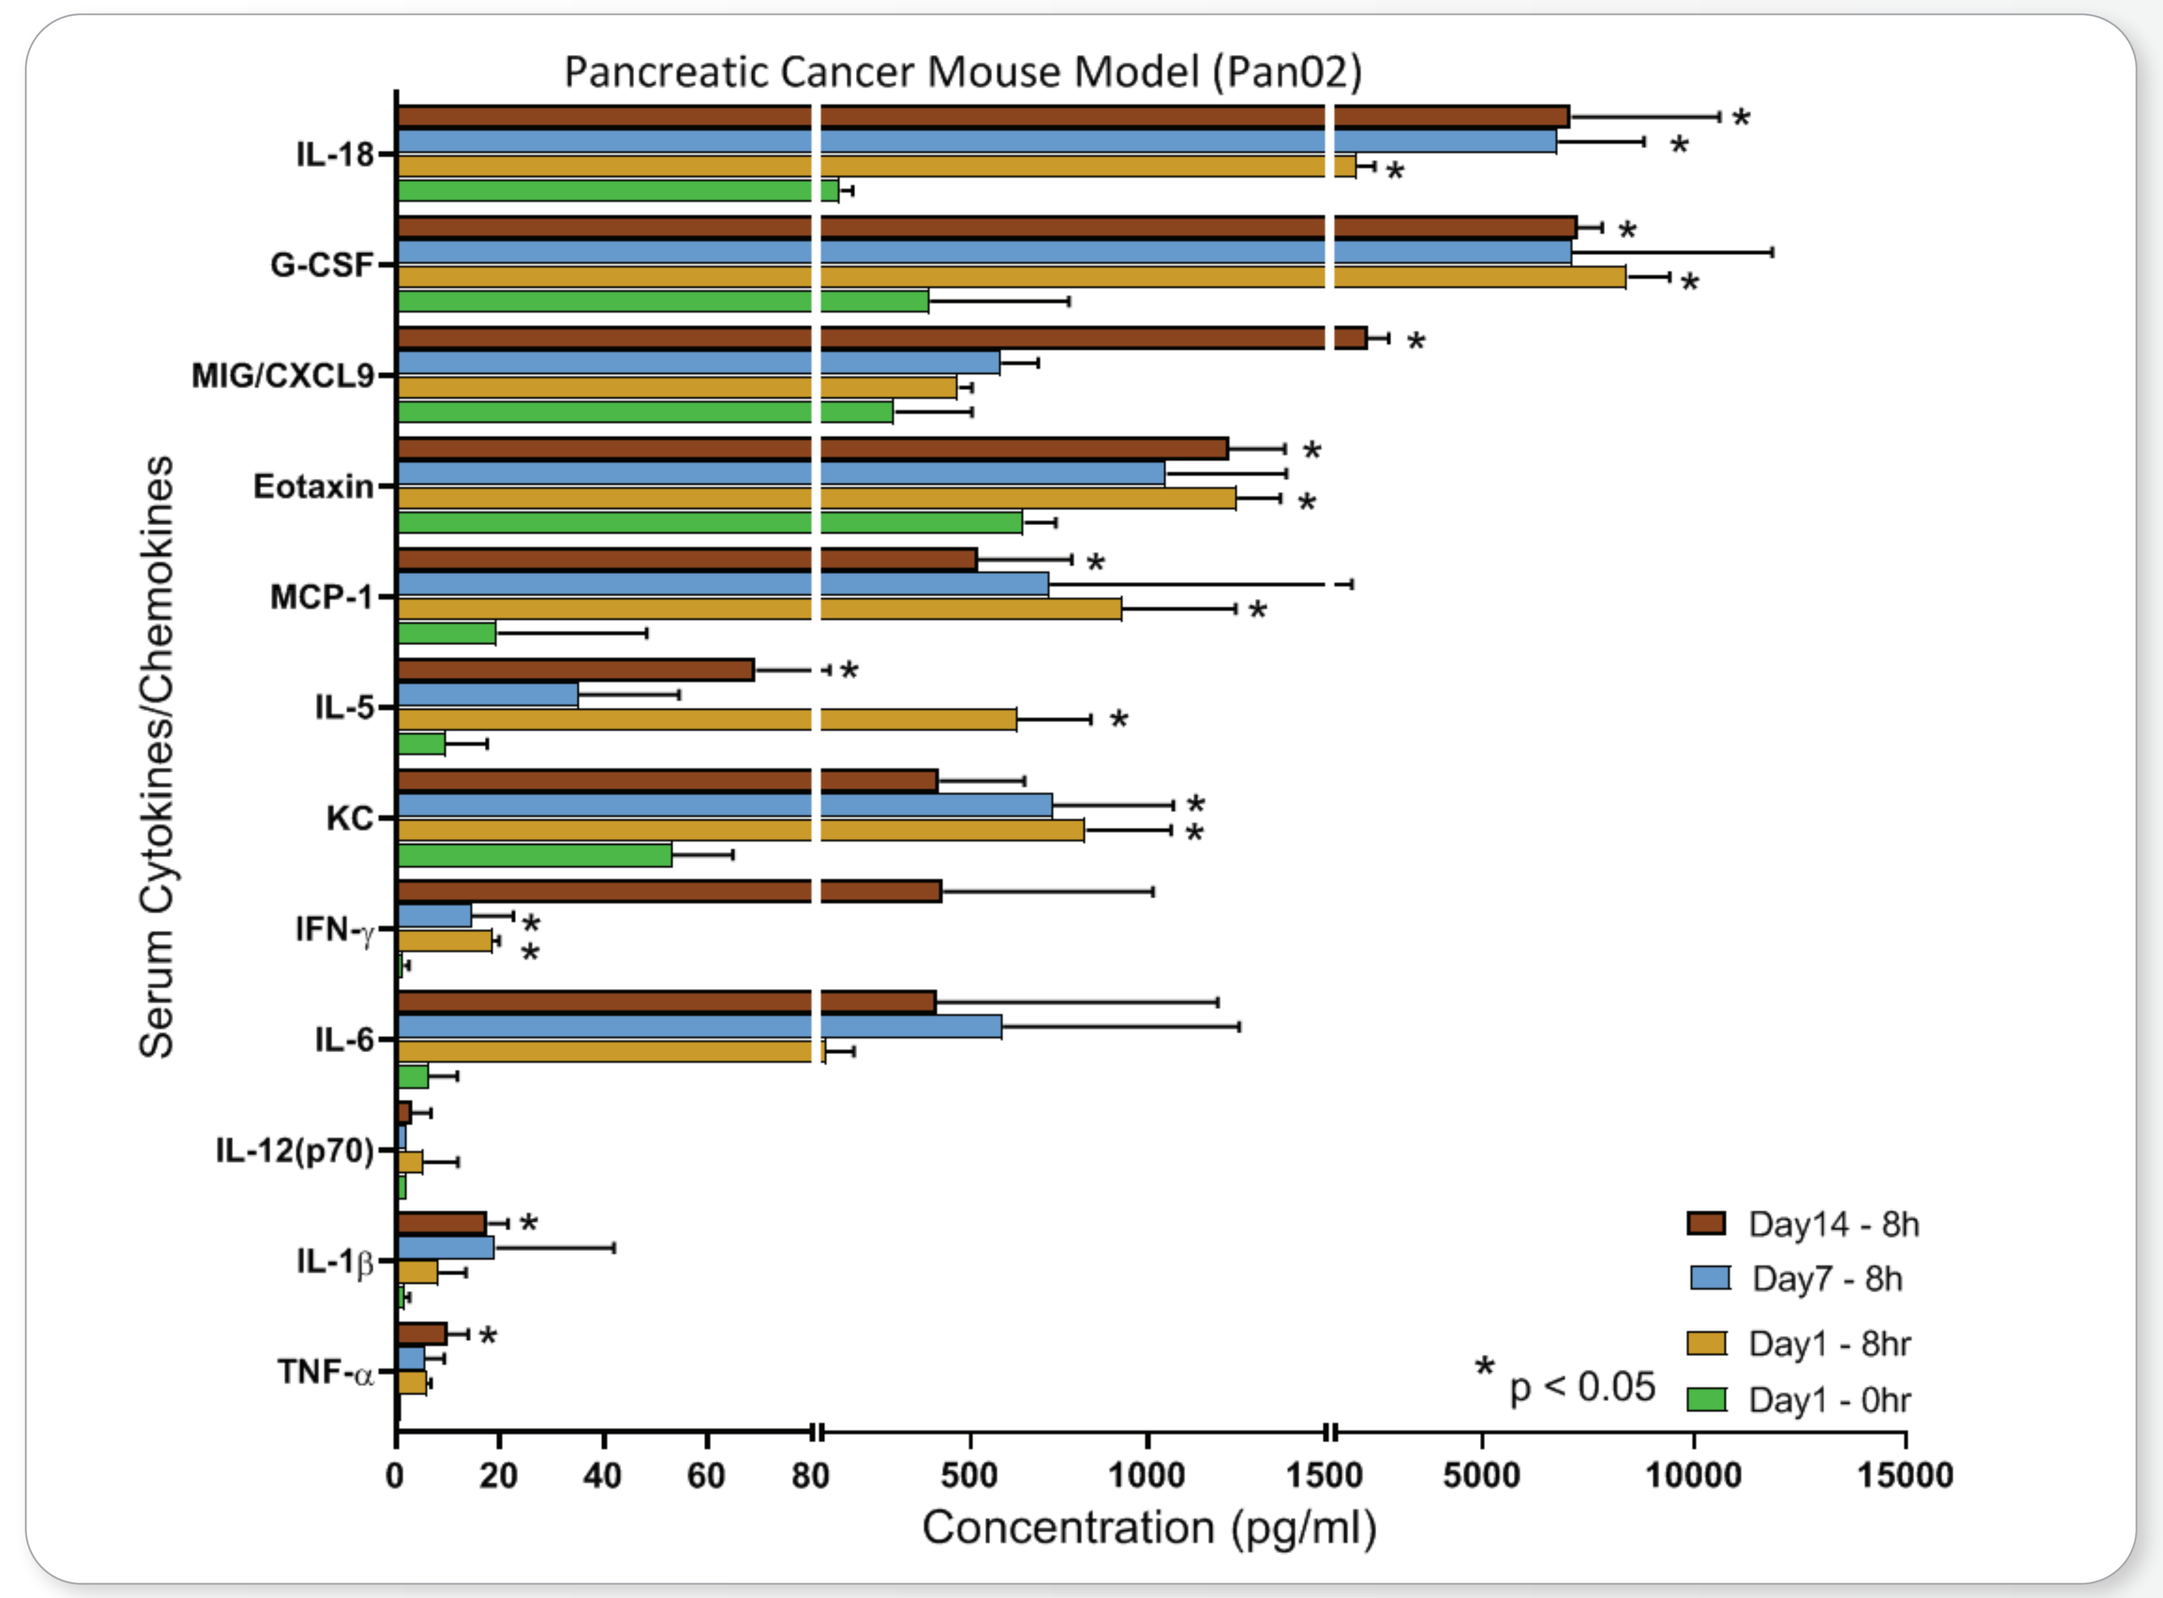
Pancreatic Cancer Mouse Model (Pan02)
Serum Cytokines/Chemokines
IL-18
G-CSF
MIG/CXCL9
Eotaxin
MCP-1
IL-5
KC
IFN-γ
IL-6
IL-12(p70)
IL-1β
TNF-α
Concentration (pg/ml)
Day14 - 8h
Day7 - 8h
Day1 - 8hr
Day1 - 0hr
* p < 0.05

Pro-inflammatory cytokines were found to be upregulated. Major upregulated cytokines were IL-18, G-CSF, MIG, Eotaxin, MCP-1, IL-5, KC, IL-6 and IFN-γ.

BXCL701 ENHANCES MHC-I EXPRESSION AND INFILTRATION OF NK CELLS AND T-CELLS IN PANCREATIC CANCER MOUSE MODEL (PAN02)

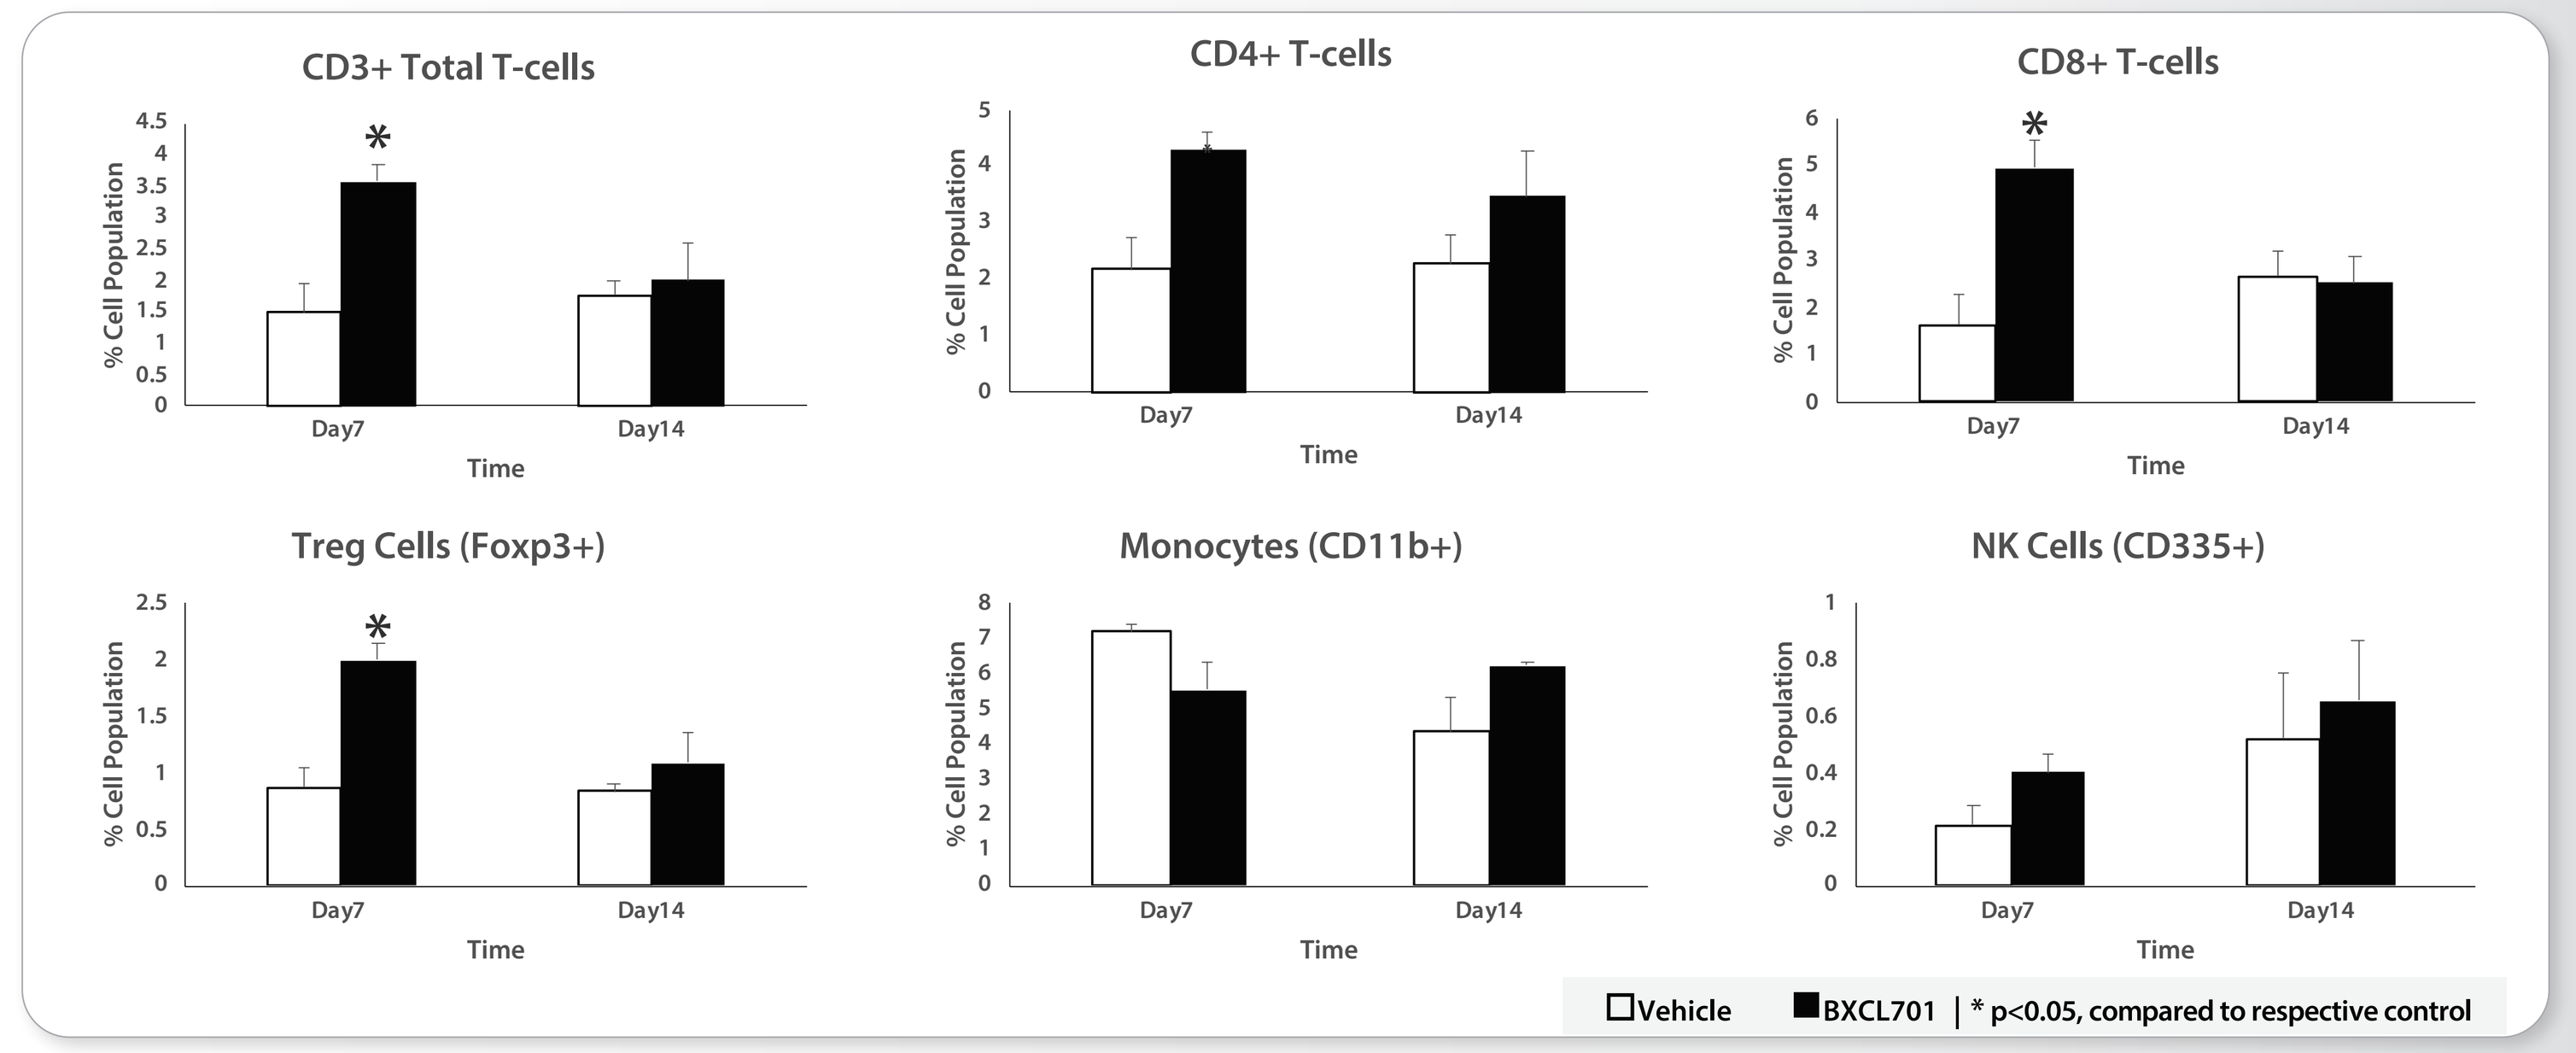
CD3+ Total T-cells
CD4+ T-cells
CD8+ T-cells
Treg Cells (Foxp3+)
Monocytes (CD11b+)
NK Cells (CD335+)
% Cell Population
Day7
Day14
Time
Vehicle
BXCL701 | * p<0.05, compared to respective control

A general increase in T lymphocyte infiltration was observed in BXCL701 treated mice as compared to vehicle treated mice. Some increase in NK cells was also observed in BXCL701 treated mice. This increase was not significant though.

Gene Expression

Differentially modulated genes with log2 fold change > 1 were selected and significantly enriched (p<0.05) gene clusters based on gene functions were reported.

GENES MODULATED BY BXCL701 IN PANCREATIC CANCER MOUSE MODEL (PAN02) ON DAY 1

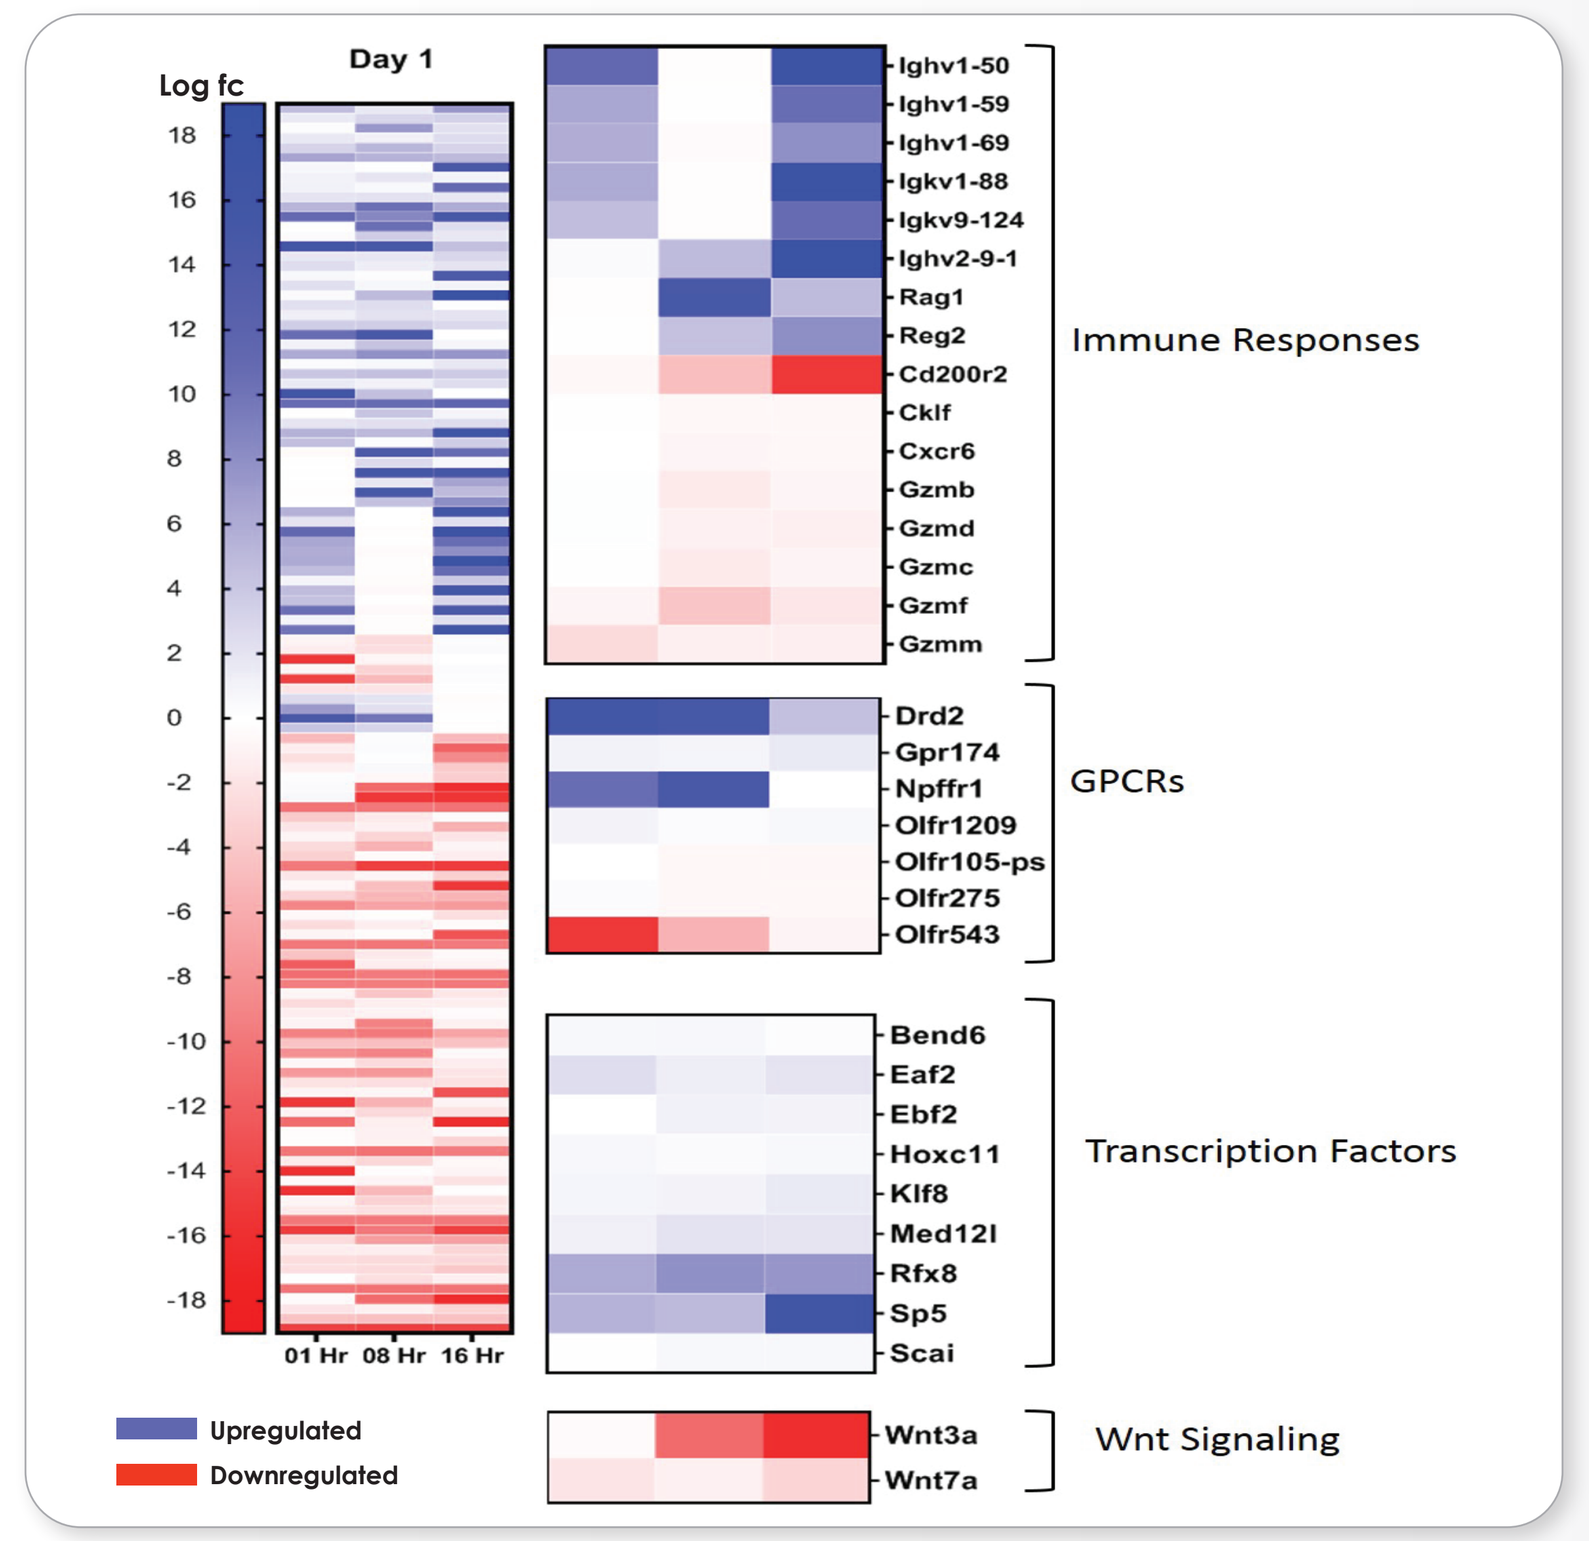
Log fc
Day 1
Immune Responses
GPCRs
Transcription Factors
Wnt Signaling
Upregulated
Downregulated

On day 1,

BXCL701 stimulates innate immune responses.

A few Ig domain containing genes, GPCRs and transcription factors involved in immune responses were found to be upregulated.

Also, Wnt signaling was observed to be downregulated.

Gene expression at Day 1 was calculated as BXCL701 T=1, 8, 16 hrs vs T=0.

GENES MODULATED BY BXCL701 IN PANCREATIC CANCER MOUSE MODEL (PAN02) ON DAY 7

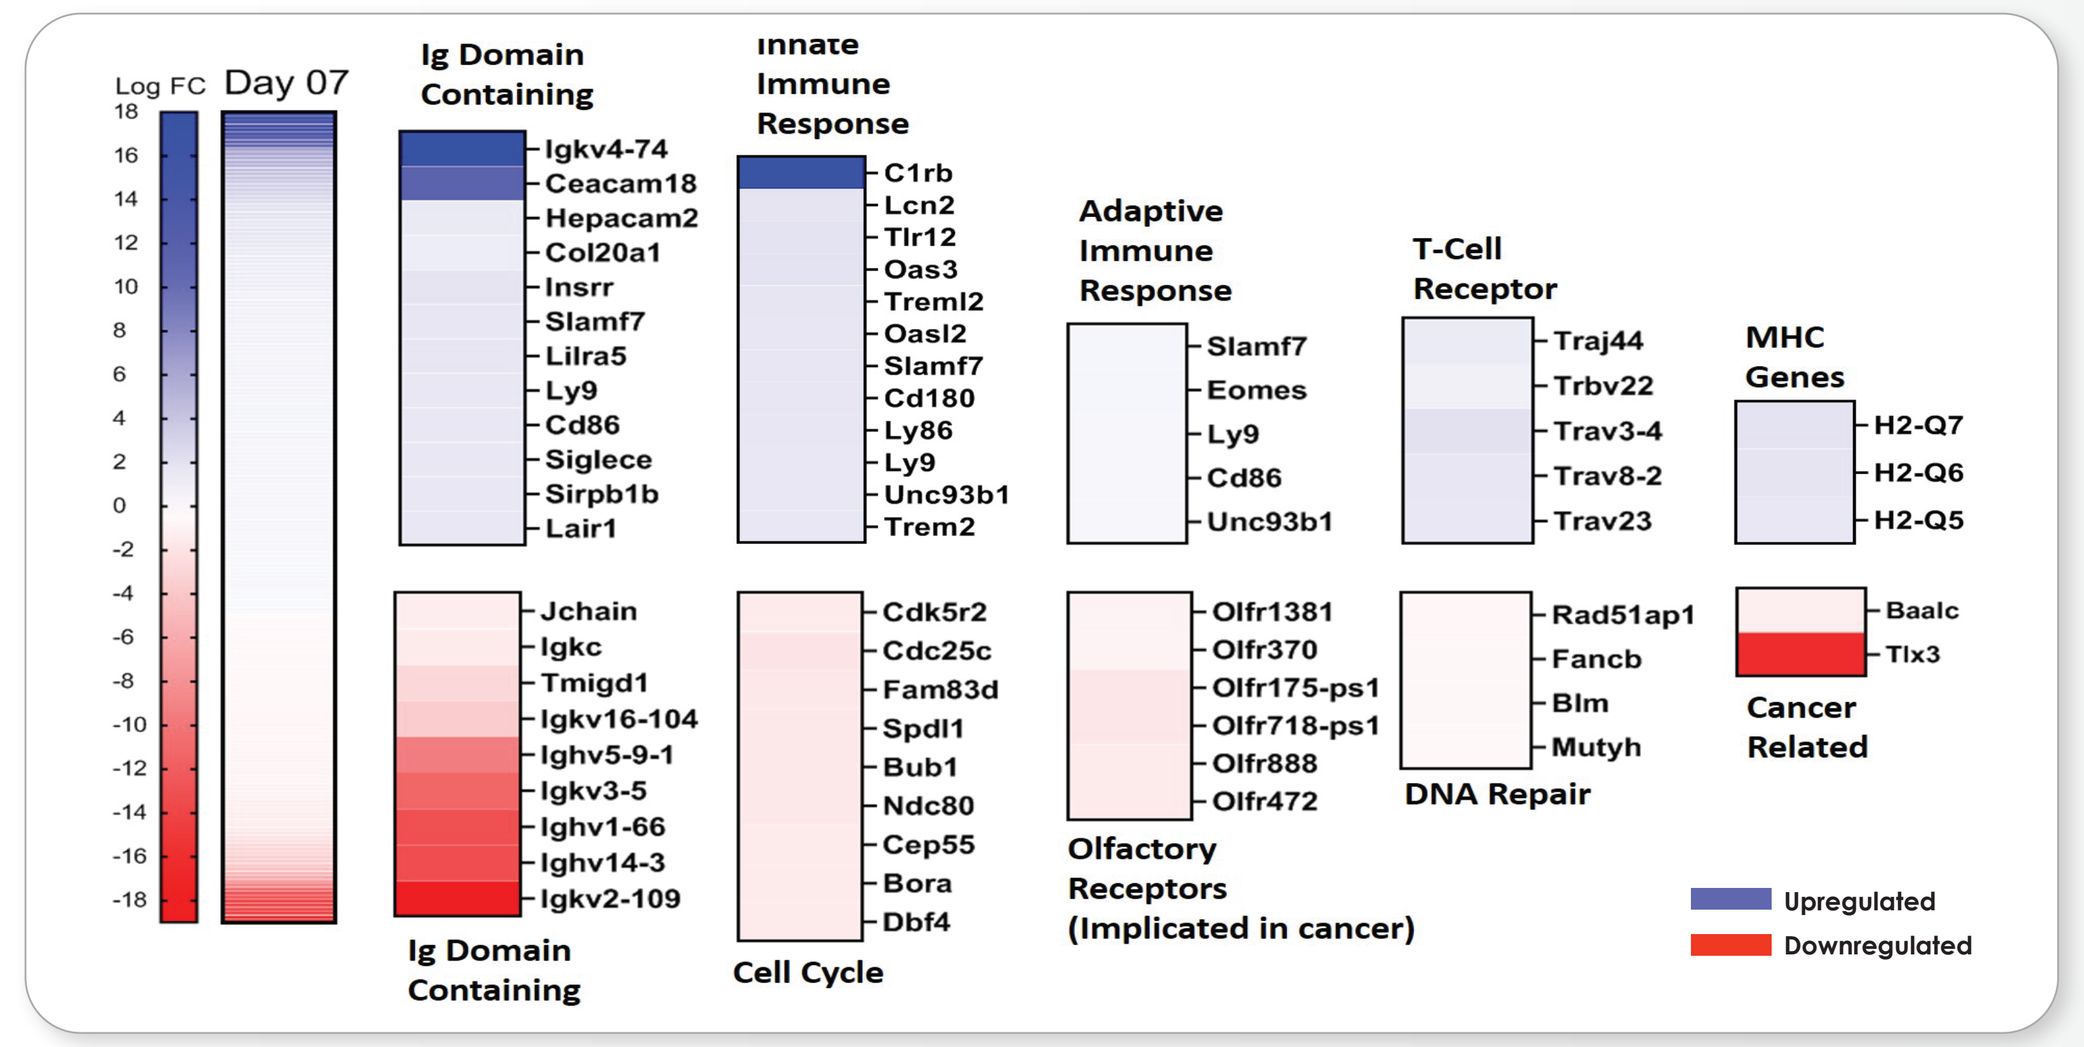
Log FC
Day 07
Ig Domain Containing
Innate Immune Response
Adaptive Immune Response
T-Cell Receptor
MHC Genes
Cancer Related
DNA Repair
Olfactory Receptors (Implicated in cancer)
Cell Cycle
Upregulated
Downregulated

By day 7, BXCL701 stimulates innate immune responses followed by adaptive immune responses. Markedly upregulated gene clusters belong to different innate immune response genes such as genes containing Immunoglobulin (Ig) Domain, MHC Genes and other genes involved in innate immune signalling.

In addition, a few adaptive immune responses that include T-cell receptor genes and genes involved in adaptive immune signalling were found to be upregulated. Downregulated gene clusters belonged to functional categories like cell cycle, DNA repair and Olfactory receptors (implicated in cancer progression). Modulation of gene expression was calculated as BXCL701, T=0 vs vehicle treated, T=0.

GENES MODULATED BY BXCL701 IN PANCREATIC CANCER MOUSE MODEL (PAN02) ON DAY 14

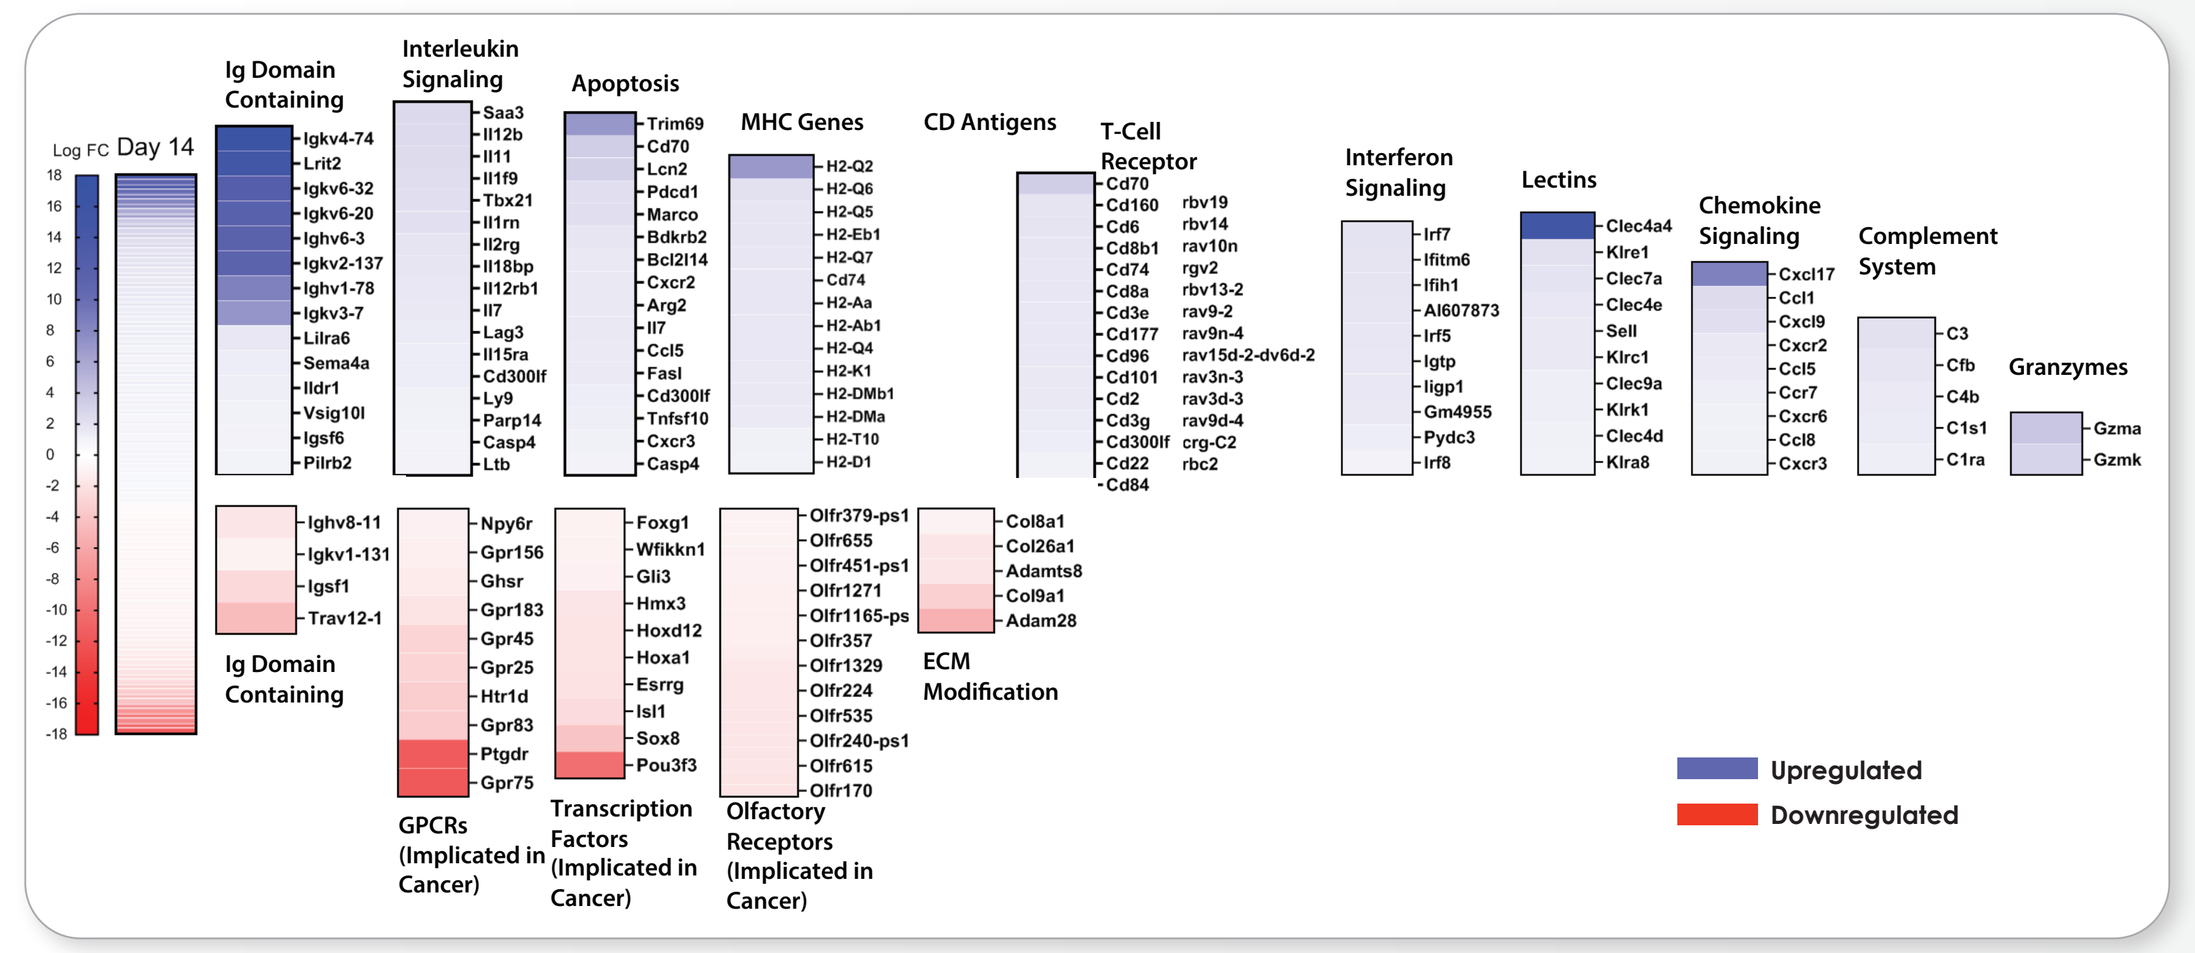
Log FC
Day 14
Ig Domain Containing
Interleukin Signaling
Apoptosis
MHC Genes
CD Antigens
T-Cell Receptor
Interferon Signaling
Lectins
Chemokine Signaling
Complement System
Granzymes
GPCRs (Implicated in Cancer)
Transcription Factors (Implicated in Cancer)
Olfactory Receptors (Implicated in Cancer)
ECM Modification
Upregulated
Downregulated

By day 14, BXCL701 Bridges Innate and Adaptive Immune Responses.
Most upregulated genes belonged to different innate immune gene groups along with a few functional clusters belonging to adaptive immunity.

Enhanced expression of lectin molecules along with Interleukin, Interferon and Chemokine signalling will lead to increase in infiltration of innate immune cells like monocytes, macrophages and NK cells which can induce tumor cell killing.

In addition, genes associated with T cell and NK cell mediated apoptosis and cytolysis e.g. FasL, GzmA, were upregulated and indicates enhanced cell death (e.g. upregulation of Casp4 within tumor. On the other hand, downregulated gene clusters belonged to functional categories like GPCRs, Transcription factors and Olfactory receptors associated with cancer progression along with extra cellular matrix (ECM) modification (collagen and metalloproteases). Gene expression at Day 14 was calculated as BXCL701, T=0 vs vehicle treated, T=0.

BXCL701 CONCENTRATION IN PLASMA AND TUMORS

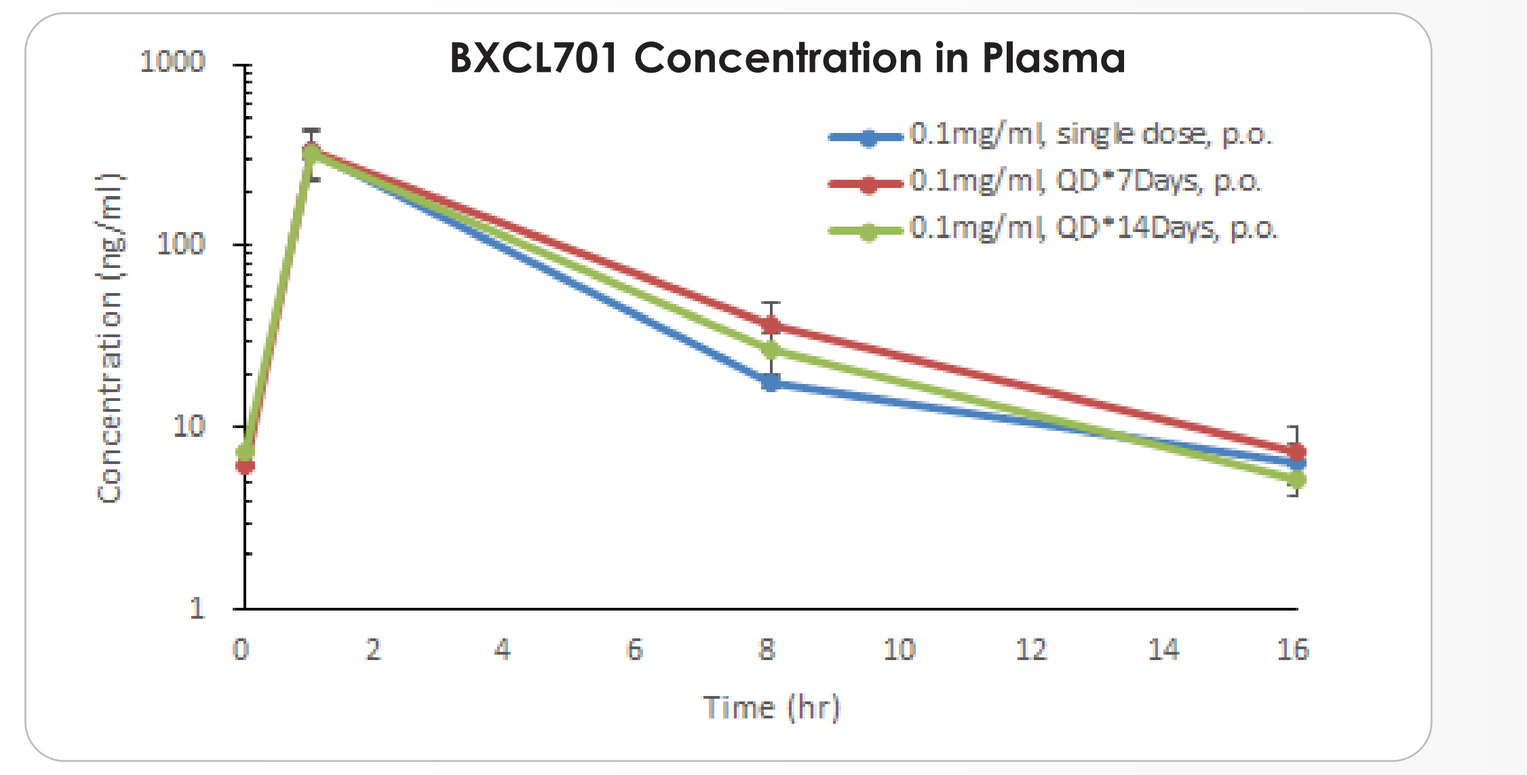
BXCL701 Concentration in Plasma
Concentration (ng/ml)
0.1mg/ml, single dose, p.o.
0.1mg/ml, QD*7Days, p.o.
0.1mg/ml, QD*14Days, p.o.
Time (hr)

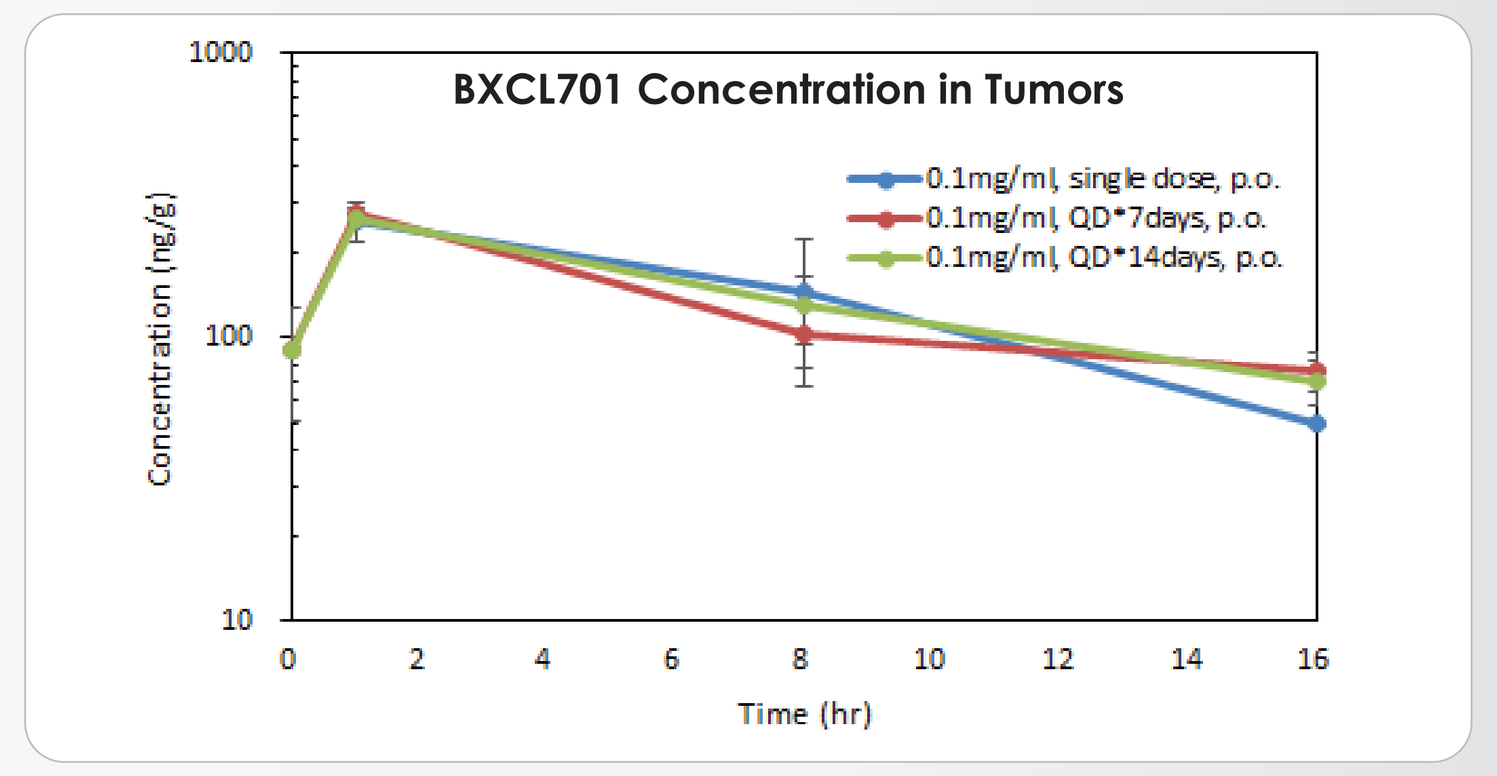
BXCL701 Concentration in Tumors
Concentration (ng/g)
0.1mg/ml, single dose, p.o.
0.1mg/ml, QD*7days, p.o.
0.1mg/ml, QD*14days, p.o.
Time (hr)

- No change in plasma/tumor concentration of BXCL701 after multiple dosing since the concentration of the molecules was almost similar at day 1, 7 and 14.
- The concentration was higher in tumor samples in comparison to plasma samples.

CONCLUSIONS

- BXCL701 bridges innate and adaptive immunity mainly via IL-18 (IL-1 family) and OX40- pathway [Agarwal et. al. 2019].
- BXCL701 enhances infiltration of T-cells and NK cells along with expression of MHC genes within the tumor microenvironment.
- The upregulated gene clusters were innate and adaptive immune response genes, T-cell receptor genes and MHC genes. In addition, genes associated with T cell and NK cell mediated apoptosis and cytolysis were upregulated and indicates enhanced cell death within tumor.
- On the other hand, downregulated gene clusters belonged to functional categories like cell cycle, DNA repair, several genes associated with cancer progression and extracellular matrix (ECM) modification.
- In conclusion, BXCL701-treatment induces innate immune responses followed by adaptive immune responses that leads to tumor growth inhibition probably via inducing cell death and reducing ECM modification.

BXCL701 MODULATES TUMOR MICROENVIRONMENT BY ACTIVATING INNATE IMMUNITY FOLLOWED BY ADAPTIVE IMMUNITY LEADING TO CELL DEATH

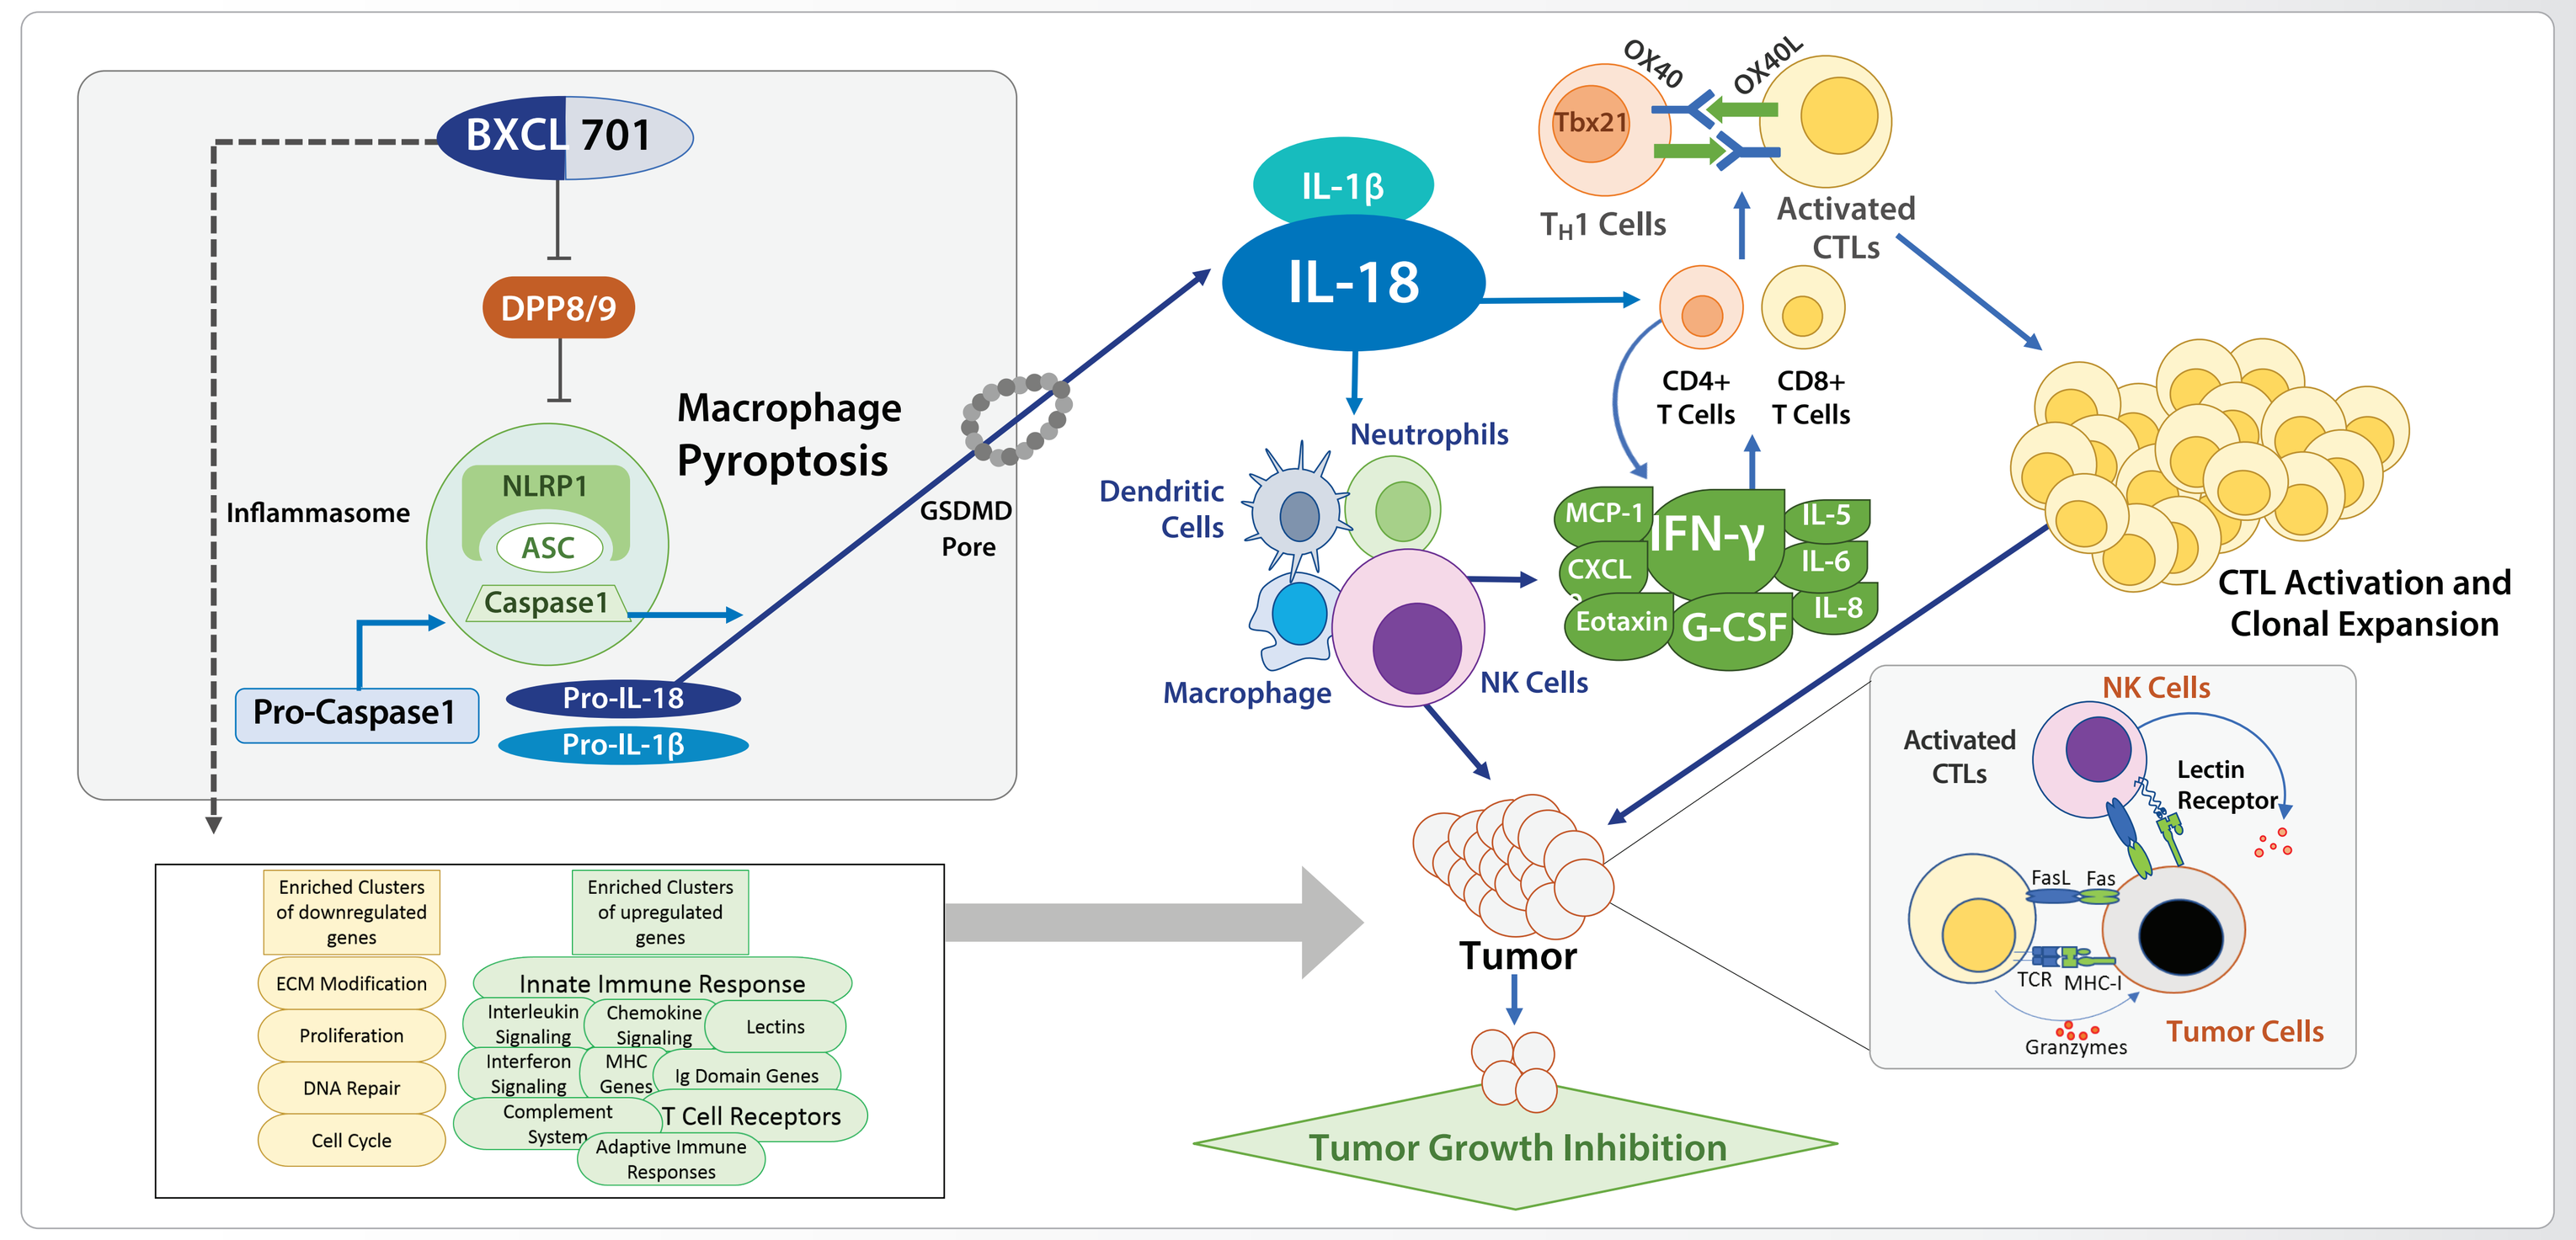
BXCL 701
DPP8/9
Inflammasome
NLRP1
ASC
Caspase1
Macrophage Pyroptosis
GSDMD Pore
Pro-Caspase1
Pro-IL-18
Pro-IL-1β
IL-1β
IL-18
Tbx21
OX40
OX40L
TH1 Cells
Activated CTLs
CD4+ T Cells
CD8+ T Cells
Neutrophils
Dendritic Cells
Macrophage
NK Cells
MCP-1
CXCL
Eotaxin
IFN-γ
G-CSF
IL-5
IL-6
IL-8
CTL Activation and Clonal Expansion
Enriched Clusters of downregulated genes
ECM Modification
Proliferation
DNA Repair
Cell Cycle
Enriched Clusters of upregulated genes
Innate Immune Response
Interleukin Signaling
Chemokine Signaling
Lectins
Interferon Signaling
MHC Genes
Ig Domain Genes
Complement System
T Cell Receptors
Adaptive Immune Responses
Tumor
Tumor Growth Inhibition
Activated CTLs
Lectin Receptor
FasL
Fas
TCR
MHC-I
Granzymes
Tumor Cells

REFERENCES

[1] Rastelli et al., 2018 ACSO Annual Meeting, Abstract #3085
[2] Okondo et al. 2018, Cell Chem. Biol. 25, 1-6
[3] Bergsbaken et al. 2009, Nat Rev Microbiol. 7(2): 99–109
[4] Agarwal et al. 2019 AACR Annual Meeting, Abstract # LB-077

ACKNOWLEDGMENTS

We would like to Acknowledge Rita Ren (in life support of animal studies), Xiaolong Tu (IHC and Cytokines evaluation), Bonnie Chen (mRNA isolation and RNASeq experiments) and all of the team members at Crown Bioscience 11011 Torreyana Road, Suite 200, San Diego, CA 92121 for running the studies.

American Association of Cancer Research- Virtual Meeting II, June 22-24, 2020, Presenter's contact information: Veena Agarwal, Ph.D., Email ID: vagarwal@bioxceltherapeutics.com