Fluorocyclopentenylcytosine (RX-3117) is activated by uridine-cytidine kinase 2, a potential biomarker

Godefridus J. Peters[1], Joris R Julsing[1], Kees Smid[1], Daniel de Klerk[1], Dzjemma Sarkisjan[1], Mi Young Yang[2], Young Bok Lee[2], Deog Joong Kim[2],

[1]Department of Medical Oncology, VU University Medical Center, Amsterdam, The Netherlands; [2]Rexahn Pharmaceuticals, Inc., Rockville, MD, USA

Poster # 2622 AACR Annual Meeting 2015

INTRODUCTION

- RX-3117 (fluorocyclopentenylcytosine) is a novel cytidine analog[1]
- RX-3117 resembles ethynylcytidine (Ecyd) and azacytidine (aza-CR)
- RX-3117 is incorporated into RNA and DNA

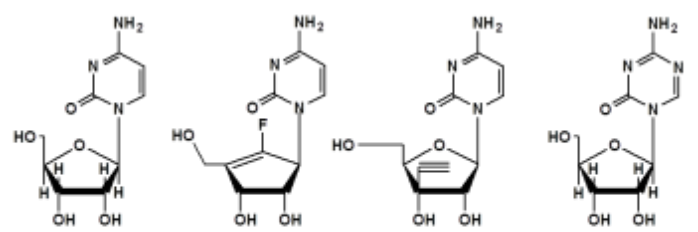

Cytidine **RX-3117** **ECyd** **Aza-CR**

- RX-3117 is taken up by the equilibrative nucleoside transporter (ENT)
- Preliminary data indicated that it inhibits DNA methyltransferase (DNMT) [1,2]
- RX-3117 is active in cell lines and tumors resistant to gemcitabine[2,3]
- RX-3117 is metabolized by an uridine-cytidine kinase (UCK)
- There are two forms: UCK1 and UCK2

AIMS OF THE STUDY

1. **Is RX-3117 activated by UCK1 or UCK2?**
2. **What is the expression and activity of UCK1 and UCK2 in model systems?**
3. **Can UCK2 be detected by immunohistochemistry?**
4. **Is UCK activity related to RX-3117's efficacy?**

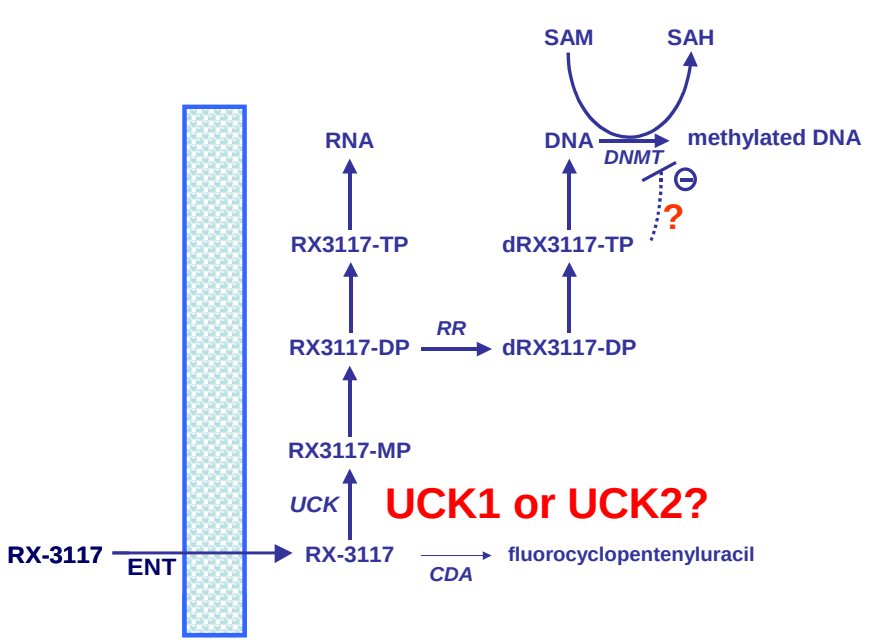

SAM
SAH
RNA
DNA
methylated DNA
DNMT
?
RX3117-TP
dRX3117-TP
RR
RX3117-DP
dRX3117-DP
RX3117-MP
UCK
UCK1 or UCK2?
RX-3117
ENT
RX-3117
CDA
fluorocyclopentenyluracil

MODEL SYSTEMS

In vitro models:
- A549 and SW1573 non-small cell lung cancer (NSCLC) cell lines
- Cells were cultured in DMEM plus 10% fetal bovine serum
- A panel of various cell lines with different UCK activity

In vivo models:
- a panel of cells from various xenografts
- 3 xenograft models with different sensitivity to RX-3117 and UCK activity

RESULTS

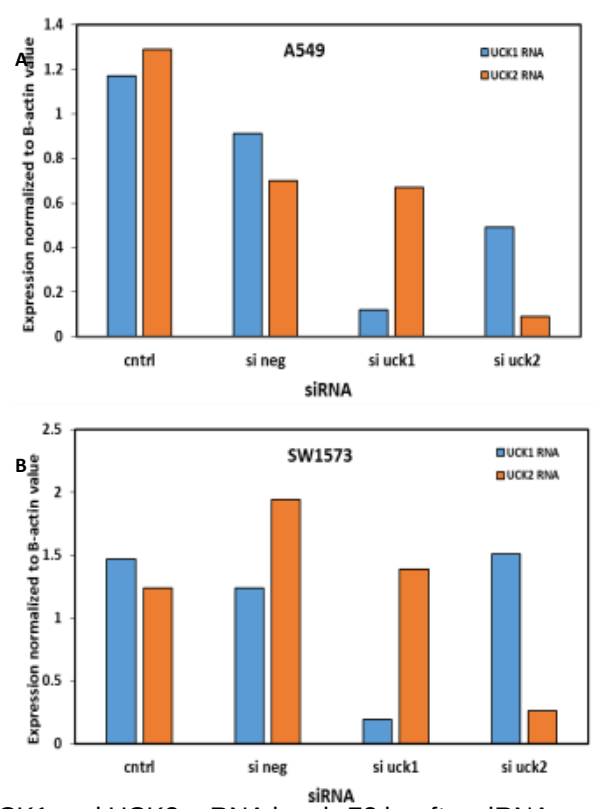

A
A549
UCK1 RNA
UCK2 RNA
Expression normalized to B-actin value
cntrl
si neg
si uck1
si uck2
siRNA
B
SW1573
UCK1 RNA
UCK2 RNA
Expression normalized to B-actin value
cntrl
si neg
si uck1
si uck2
siRNA

UCK1 and UCK2 mRNA levels 72 hr after siRNA transfection. **(A)** UCK1 and UCK2 mRNA levels after transfection in A549 cells. **(B)** UCK1 and UCK2 mRNA levels after transfection in SW1573 cells. Values are from one representative experiment of multiple performed experiments.

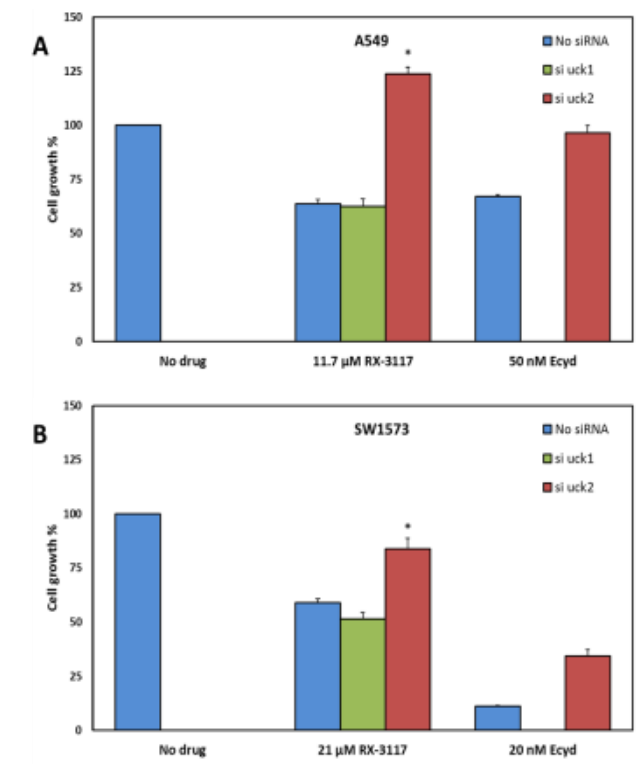

A
A549
No siRNA
si uck1
si uck2
Cell growth %
No drug
11.7 µM RX-3117
50 nM Ecyd
B
SW1573
No siRNA
si uck1
si uck2
Cell growth %
No drug
21 µM RX-3117
20 nM Ecyd

Effect of UCK1 and UCK2 down regulation on RX-3117 cytotoxicity in A549 **(A)** and SW1573 **(B)** cells. Values are means ± SEM (n=6). Values were normalized to 100% for untreated cells. Drugs were added 24 hr after transfection. Cells were exposed to drugs for 48 hr. ECyd was used as a positive control. The protective effect of siUCK2 was considered significant in both A549 (p=0.004) and SW1573 cells (p=0.003). Significance is marked with an asterisk (*).

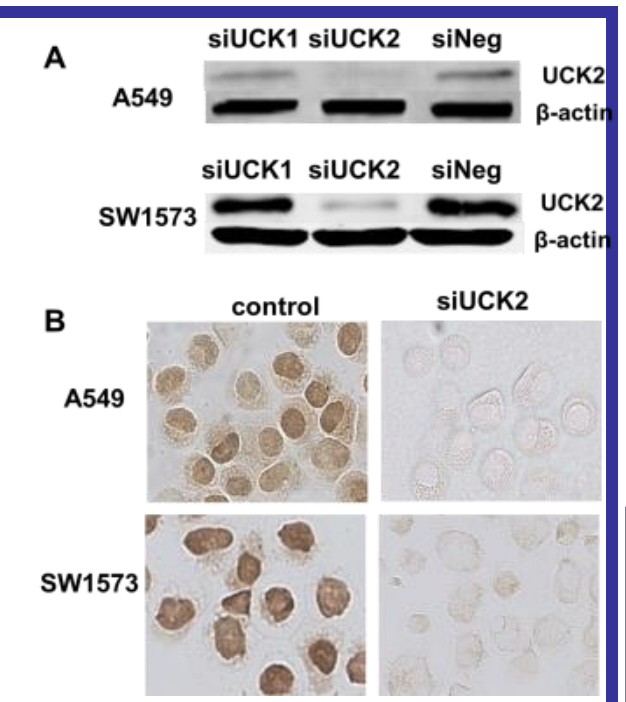

A
siUCK1 siUCK2 siNeg
A549
UCK2
β-actin
siUCK1 siUCK2 siNeg
SW1573
UCK2
β-actin
B
control
siUCK2
A549
SW1573

Validation of UCK2 down regulation. Silencing of UCK2, measured by western blotting **(A)** and immunocytochemistry **(B)**.

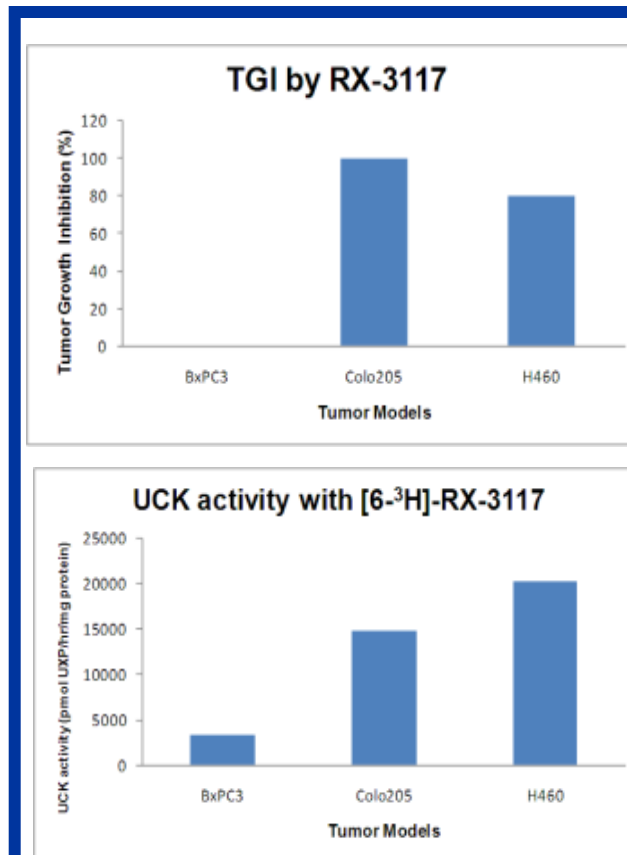

TGI by RX-3117
Tumor Growth Inhibition (%)
BxPC3
Colo205
H460
Tumor Models
UCK activity with [6-3H]-RX-3117
UCK activity (pmol UXP/hr/mg protein)
BxPC3
Colo205
H460
Tumor Models

Antitumor effect of RX-3117 (upper panel) represented as TGI (total growth inhibition in xenografts of pancreatic cancer (BXPC3), colon cancer (Colo205) and lung cancer (NCI-H460). The insensitive BxPC3 had the lowest enzyme activity (lowest panel).[3]

METHODS

- UCK activity was measured in 8000g supernatants from cell extracts and tumor homogenates by using radioactive [^{3}H]-RX-3117 and [2-^{14}C]-uridine[2]
- RNA was isolated by Trizol, reverse transcribed to cDNA
- UCK1 and 2 expression were measured by qRT-PCR using a Light-Cycler 2.0
- siRNA against UCK1 and UCK2 were obtained from Dharmacon
- Cells were transfected using Dharmafect
- Cytotoxicity to RX-3117 and Ecyd was measured by adding the drugs 24 hr after siRNA treatment, exposed to the drugs for 48 h and cytotoxicity was evaluated by SRB assays[4]
- Immunocytochemistry was measured a polyclonal rabbit antibody (YK-582) (1:200 dilution) and visualized by DAB staining.
- For Western blots, the same antibody (1:1000) was used and visualized using goat anti-rabbit InfraRedDye using an Odyssey InfraRed imager.
- Statistics were done using the Student's t-test and Pearson correlation.

Correspondence: gj.peters@vumc.nl

For further information about RX-3117 and Rexahn Pharmaceuticals, Inc., please contact Dr. DJ Kim:
kimdj@rexahn.com, (240) 268-5300 X 306

CONCLUSIONS

- **RX-3117 is activated by UCK2**
- **UCK2 can be detected by a specific UCK2 antibody**
- **In tumor tissues UCK2 expression relates to UCK activity**
- **RX-3117 has a high antitumor activity in tumors with a high UCK activity**

RESULTS

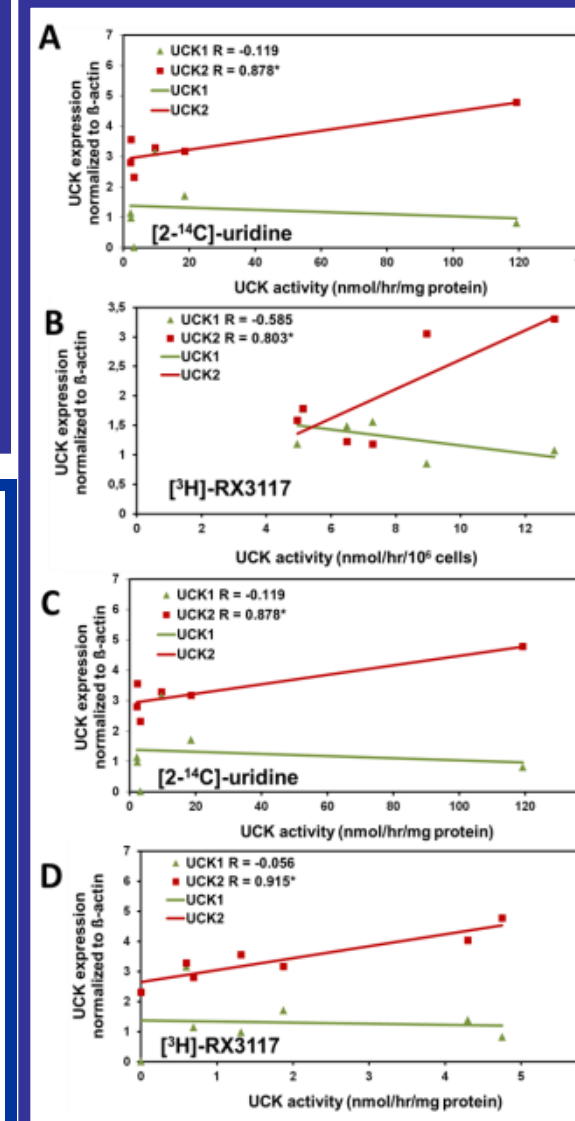

A
UCK1 R = -0.119
UCK2 R = 0.878*
UCK1
UCK2
UCK expression normalized to ß-actin
[2-14C]-uridine
UCK activity (nmol/hr/mg protein)
B
UCK1 R = -0.585
UCK2 R = 0.803*
UCK1
UCK2
UCK expression normalized to ß-actin
[3H]-RX3117
UCK activity (nmol/hr/10^6 cells)
C
UCK1 R = -0.119
UCK2 R = 0.878*
UCK1
UCK2
UCK expression normalized to ß-actin
[2-14C]-uridine
UCK activity (nmol/hr/mg protein)
D
UCK1 R = -0.056
UCK2 R = 0.915*
UCK1
UCK2
UCK expression normalized to ß-actin
[3H]-RX3117
UCK activity (nmol/hr/mg protein)

Regression analysis of UCK1/UCK2 expression and enzyme activity for [2-^{14}C]-uridine **(A)** and [^{3}H]-RX-3117 **(B)** in the cell line panel and xenograft cells (**C** and **D**). In both panels UCK2 mRNA expression correlated significantly (p<0.001) with UCK enzyme activity measured with both [2-^{14}C]-uridine (r=0.828 for the cell lines and r=0.878 for the xenograft cells) and [^{3}H]-RX-3117 (r=0.803 for the cell lines and r=0.915 for the xenograft cells). UCK1 mRNA expression did not correlate with enzyme activity with either substrate (r=-0.721 and -0.585, respectively for the cell lines and r=-0.119 and r=-0.056 respectively for the xenograft cells). The r-coefficient is given in the legend along for each gene. Significant correlations are marked with an asterisk (*).

References

1. Choi W.J., et al,, J. Med Chem, 55 (2012) 4521-4525.
2. Peters G.J., et al. Invest New Drugs, 31 (2013) 1444-1457
3. Yang M.Y et al. Anticancer Research, 34 (2014) 6951-6959.
4. Keepers Y.P., et al Eur J Cancer, 27 (1991) 897-900