A PHASE 1/2 STUDY OF RX-3117, AN ORAL ANTIMETABOLITE NUCLEOSIDE, IN COMBINATION WITH NAB-PACLITAXEL (NAB-PAC) AS FIRST LINE TREATMENT OF METASTATIC PANCREATIC CANCER (MET-PC): PRELIMINARY RESULTS

Hani M. Babiker[1], Peter N. Schlegel[2], Lee G. Hicks[3], Andrea Bullock[4], Nafisa J. Burhani[5], Ely Benaim[6], Callie Heaton[6], Christine Peterson[6], Allyson J. Ocean[7]
[1]University of Arizona Cancer Center, Tucson, AZ, USA; [2]Cancer Care Northwest, Spokane, WA, USA; [3]Baptist Health Lexington, Lexington, KY, USA; [4]Beth Israel Deaconess Medical Center, Boston, MA, USA;
[5]Joliet Oncology-Hematology Associates Ltd., Joliet, IL, USA; [6]Rexahn Pharmaceuticals, Inc., Rockville, MD, USA; [7]New York Presbyterian-Weill Cornell Medical Center, New York, NY, USA.

Abstract #420

Background: RX-3117 is an oral small molecule antimetabolite, cyclopentyl pyrimidyl nucleoside that is activated by cancer-enriched uridine cytidine kinase 2. Single agent RX-3117 has demonstrated efficacy in a phase 1/2 clinical study of RX-3117 in met-PC and bladder cancers. RX-3117 in combination with nab-pac is being evaluated as first line treatment of met-PC cancer.

Methods: This is a multicenter, open label phase 1/2 study (NCT03189914). Eligible subjects (aged ≥ 18 years) have histologically or radiologically proven met-PC with no prior therapies for metastatic disease, ECOG PS 0-1, and normal lab values. Phase 1 identified the MTD dose that is being further evaluated in the Phase 2: RX-3117 (700 mg administered orally once-daily for 5 consecutive days with 2 days off per week) and nab-pac (125 mg/m^2 administered once weekly) for 3 weeks with 1 week off per 4-week cycle. The Safety Committee reviewed data from Phase 1 before moving to Phase 2. The primary endpoint of Phase 2 (dose expansion) is an adequate number of responders based on PFS at 4 months and/or objective clinical response per RECIST v1.1.

Results: As of September 21, 2018, 8 Phase 1 subjects and 13 Phase 2 subjects were enrolled and treated (9 males and 12 females, median age 67 years). The most common (≥15%) related adverse events were nausea, diarrhea, fatigue, alopecia, decreased appetite, rash, vomiting, and anemia. Fourteen subjects had at least one on-study scan (after 2 cycles). One subject experienced a complete response (CR) after 6 cycles of therapy with normalization of CA19.9 (-76%) and confirmed after 8 cycles. Three subjects exhibited a partial response (PR): two after 2 cycles (39 and 47%) and one after 4 cycles of therapy (36%). Eight subjects had stable disease for at least 2 months, and 4 subjects had PFS of at least 4 months. The disease control rate (CR+PR+SD) was 86% in evaluable subjects while the overall response rate (CR+PR) was 29%. (update in Results section)

Conclusions: RX-3117 in combination with nab-pac appears to be safe and well tolerated in subjects with met-PC. Anti-tumor activity per RECIST was observed in 12 subjects. Phase 2 of the clinical study is currently ongoing.

Introduction

The current National Comprehensive Cancer Network recommendations suggest acceptable chemotherapy combinations for met-PC patients include FOLFIRINOX and gemcitabine plus nab-paclitaxel (nab-pac). However, clinical trials are also recommended due to the modest efficacy and severe toxicity (FOLFIRINOX) with these treatments. RX-3117, an oral small molecule antimetabolite, is phosphorylated by uridine-cytidine kinase 2 (UCK2), an enzyme found in cancer cells, but relatively absent in normal cells, allowing for a targeted approach. The targeted approach may allow for maximum dosing of RX-3117 and agents used in combination, such as nab-pac, without compromising safety. Additionally, RX-3117 is inactivated by cytidine deaminase at a slow rate, allowing higher cellular concentrations and an increased likelihood of antitumor activity at the appropriate stage of the cell cycle. Efficacy and safety of RX-3117 as a single agent in metastatic pancreatic cancer has been previously reported (Chung VM, et al., J Clin Oncol 36, 2018 (suppl 4S; abstr 396)).

Study Design

Phase 1
- Safety run-in of RX-3117 + nab-pac
- Newly diagnosed met-PC
- PK
- N=6
- DLT Determination in Cycle 1
- Complete

Phase 2 – Stage 1
- Open-label study of RX-3117 + nab-pac at RP2D
- Met-PC as first line therapy
- N=10
- Complete

Phase 2 – Stage 2
- Dose Expansion open-label design of RX-3117 + nab-pac at RP2D
- Met-PC as first line therapy
- N=30
- Ongoing

The recommended phase 2 dose (RP2D) of RX-3117, in combination with nab-pac, was determined by the safety profile and dose modification to be 700 mg administered orally once-daily for 5 consecutive days with 2 days off per week and nab-pac 125 mg/m^2 administered IV once weekly for 3 weeks with 1 week off per 4-week cycle.

Baseline Characteristics (N=36)

Age (No. of yr.)		**No. of metastatic sites — no. (%)**	
Median	67	1	9 (26)
Range	45-81	2	6 (17)
Race or Ethnic Group — no. (%)		3	12 (34)
Black	3 (8)	>3	8 (23)
White	33 (92)	**Site of metastatic disease — no. (%)**	
Non-Hispanic	36 (100)	Liver	39 (72)
Sex — no. (%)		Lung	11 (20)
Female	17 (47)	Peritoneum	4 (8)
Male	19 (53)	**Level of CA19-9 — no./total no. (%)**	
ECOG score — no. (%)		Normal	9 (25)
ECOG = 0	17 (47)	ULN to <59× ULN	13 (36)
ECOG = 1	19 (53)	≥59× ULN	8 (22)

Results

Days on Study: All Evaluable Subjects

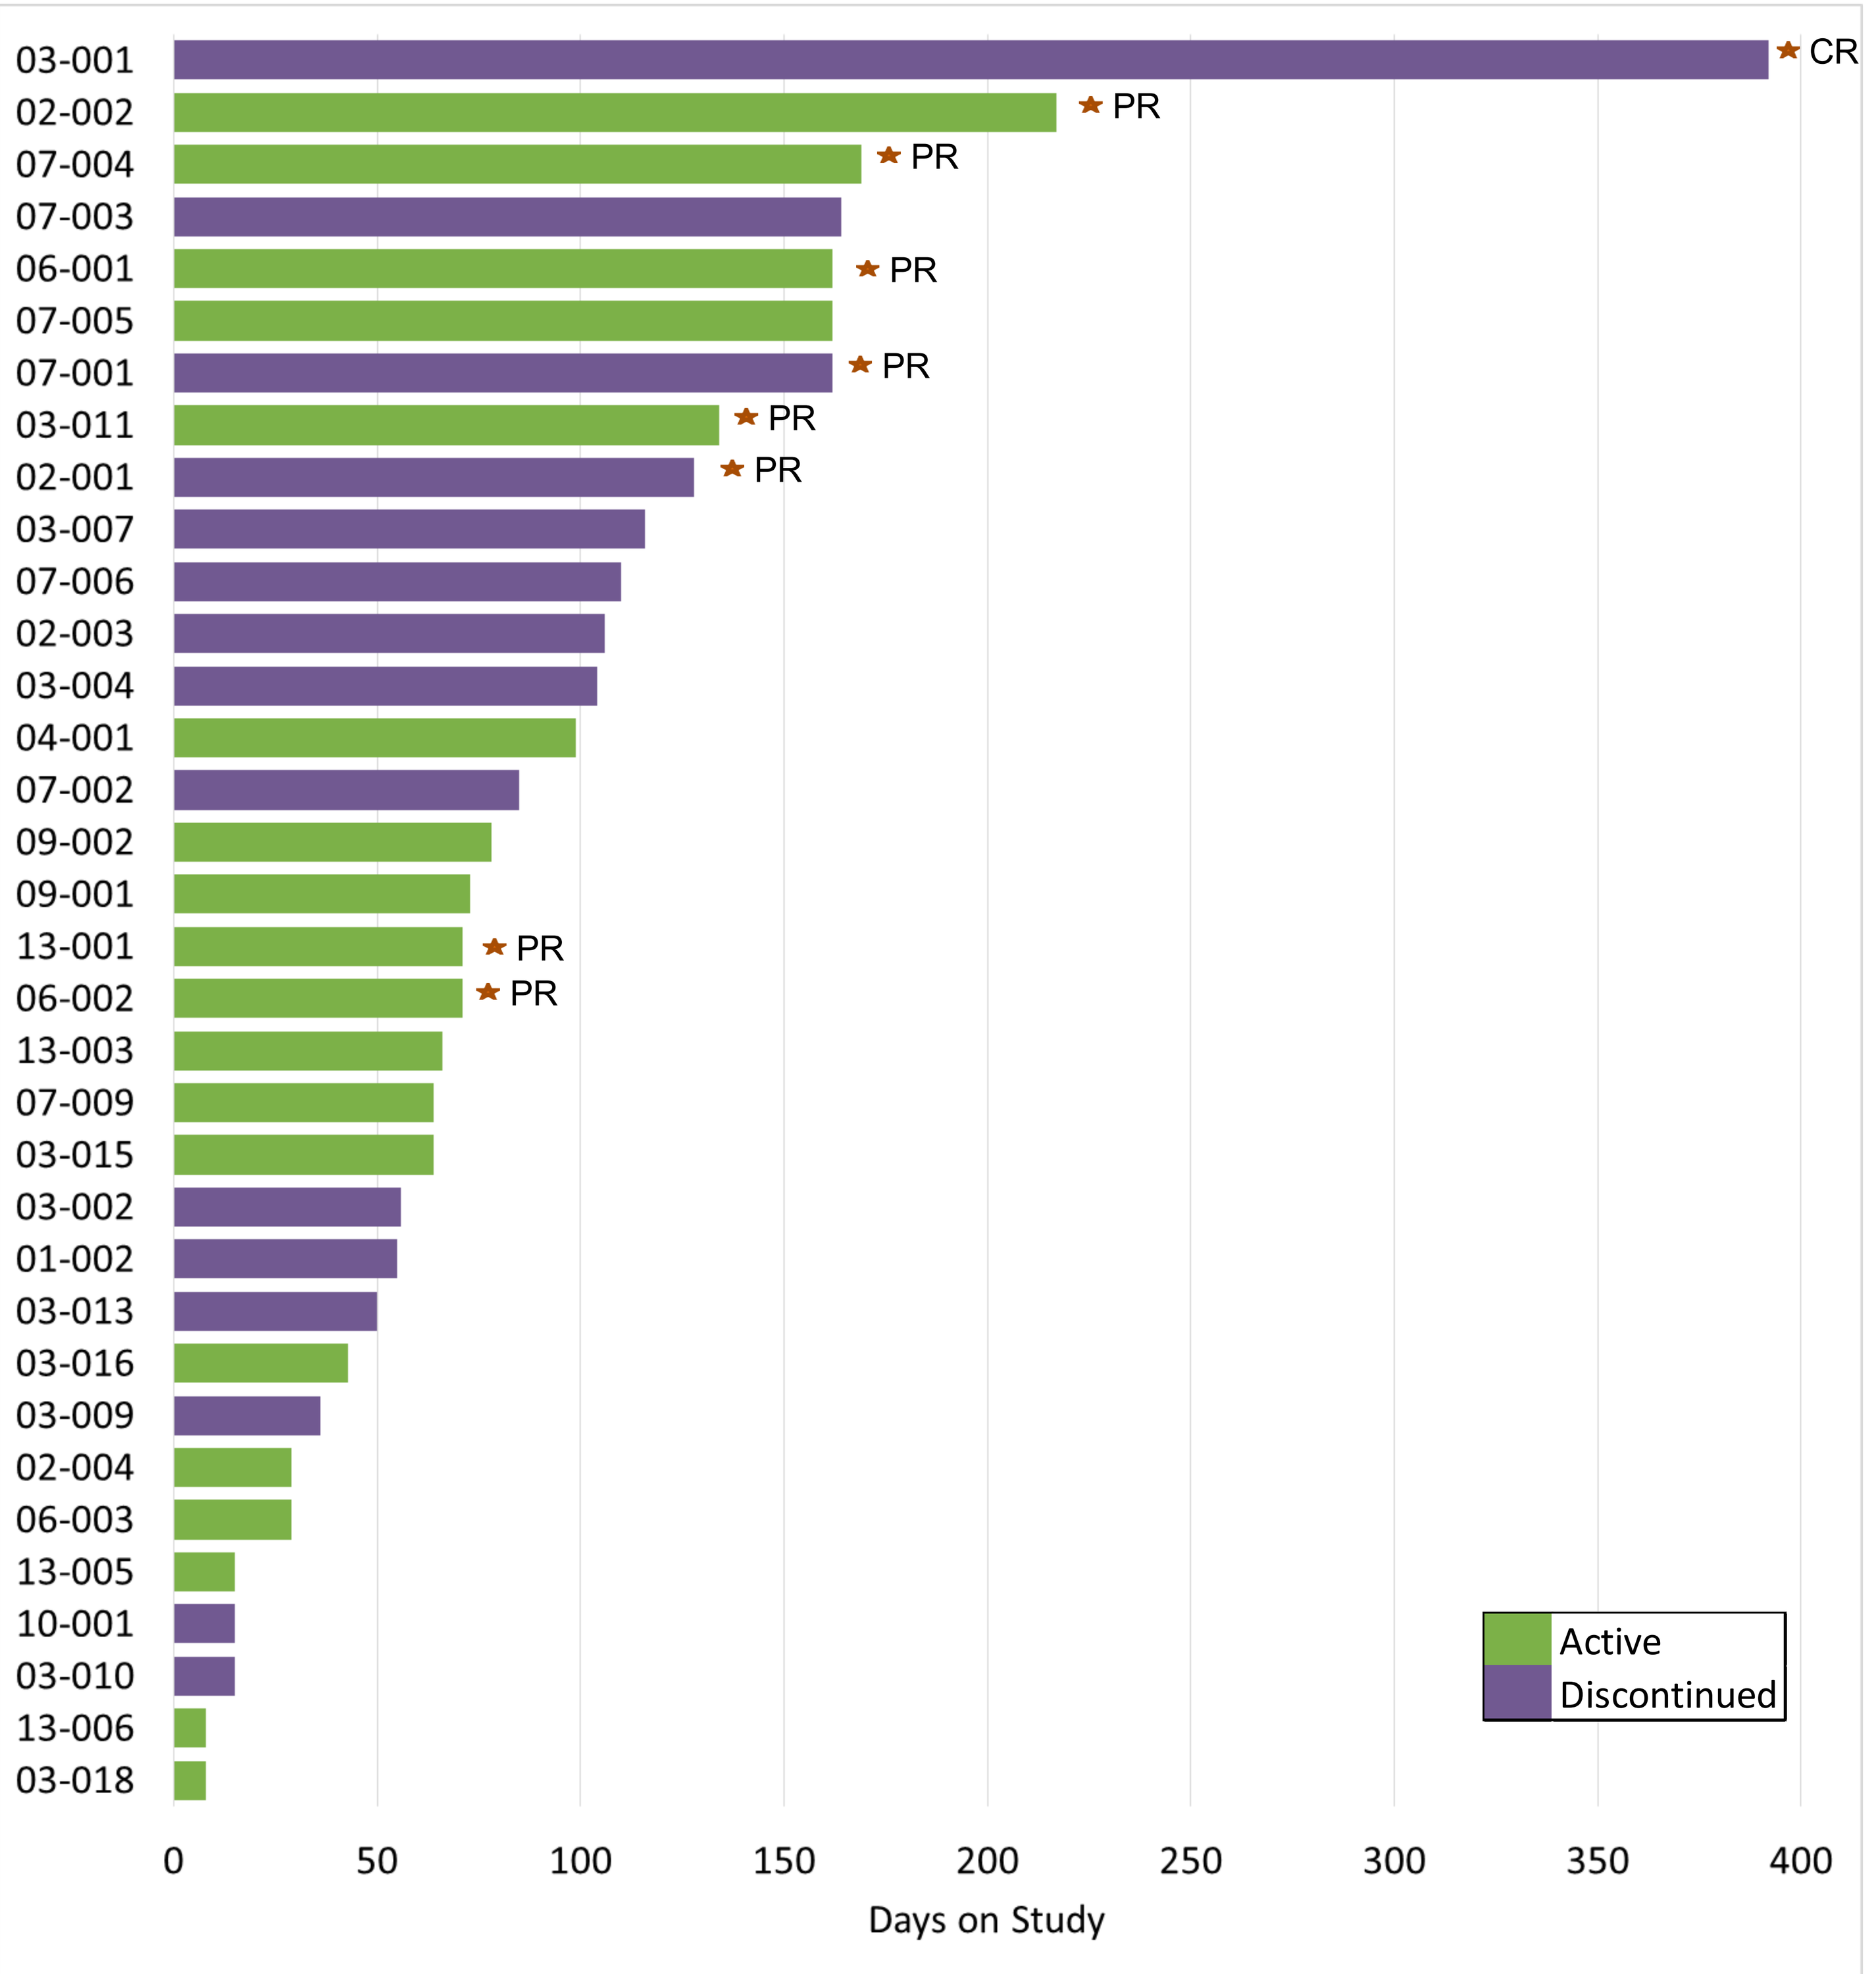
03-001 CR
02-002 PR
07-004 PR
07-003
06-001 PR
07-005
07-001 PR
03-011 PR
02-001 PR
03-007
07-006
02-003
03-004
04-001
07-002
09-002
09-001
13-001 PR
06-002 PR
13-003
07-009
03-015
03-002
01-002
03-013
03-016
03-009
02-004
06-003
13-005
10-001
03-010
13-006
03-018
0
50
100
150
200
250
300
350
400
Days on Study
Active
Discontinued

Best Overall Response per Subject by Visit (All Phases) (RECIST v1.1)

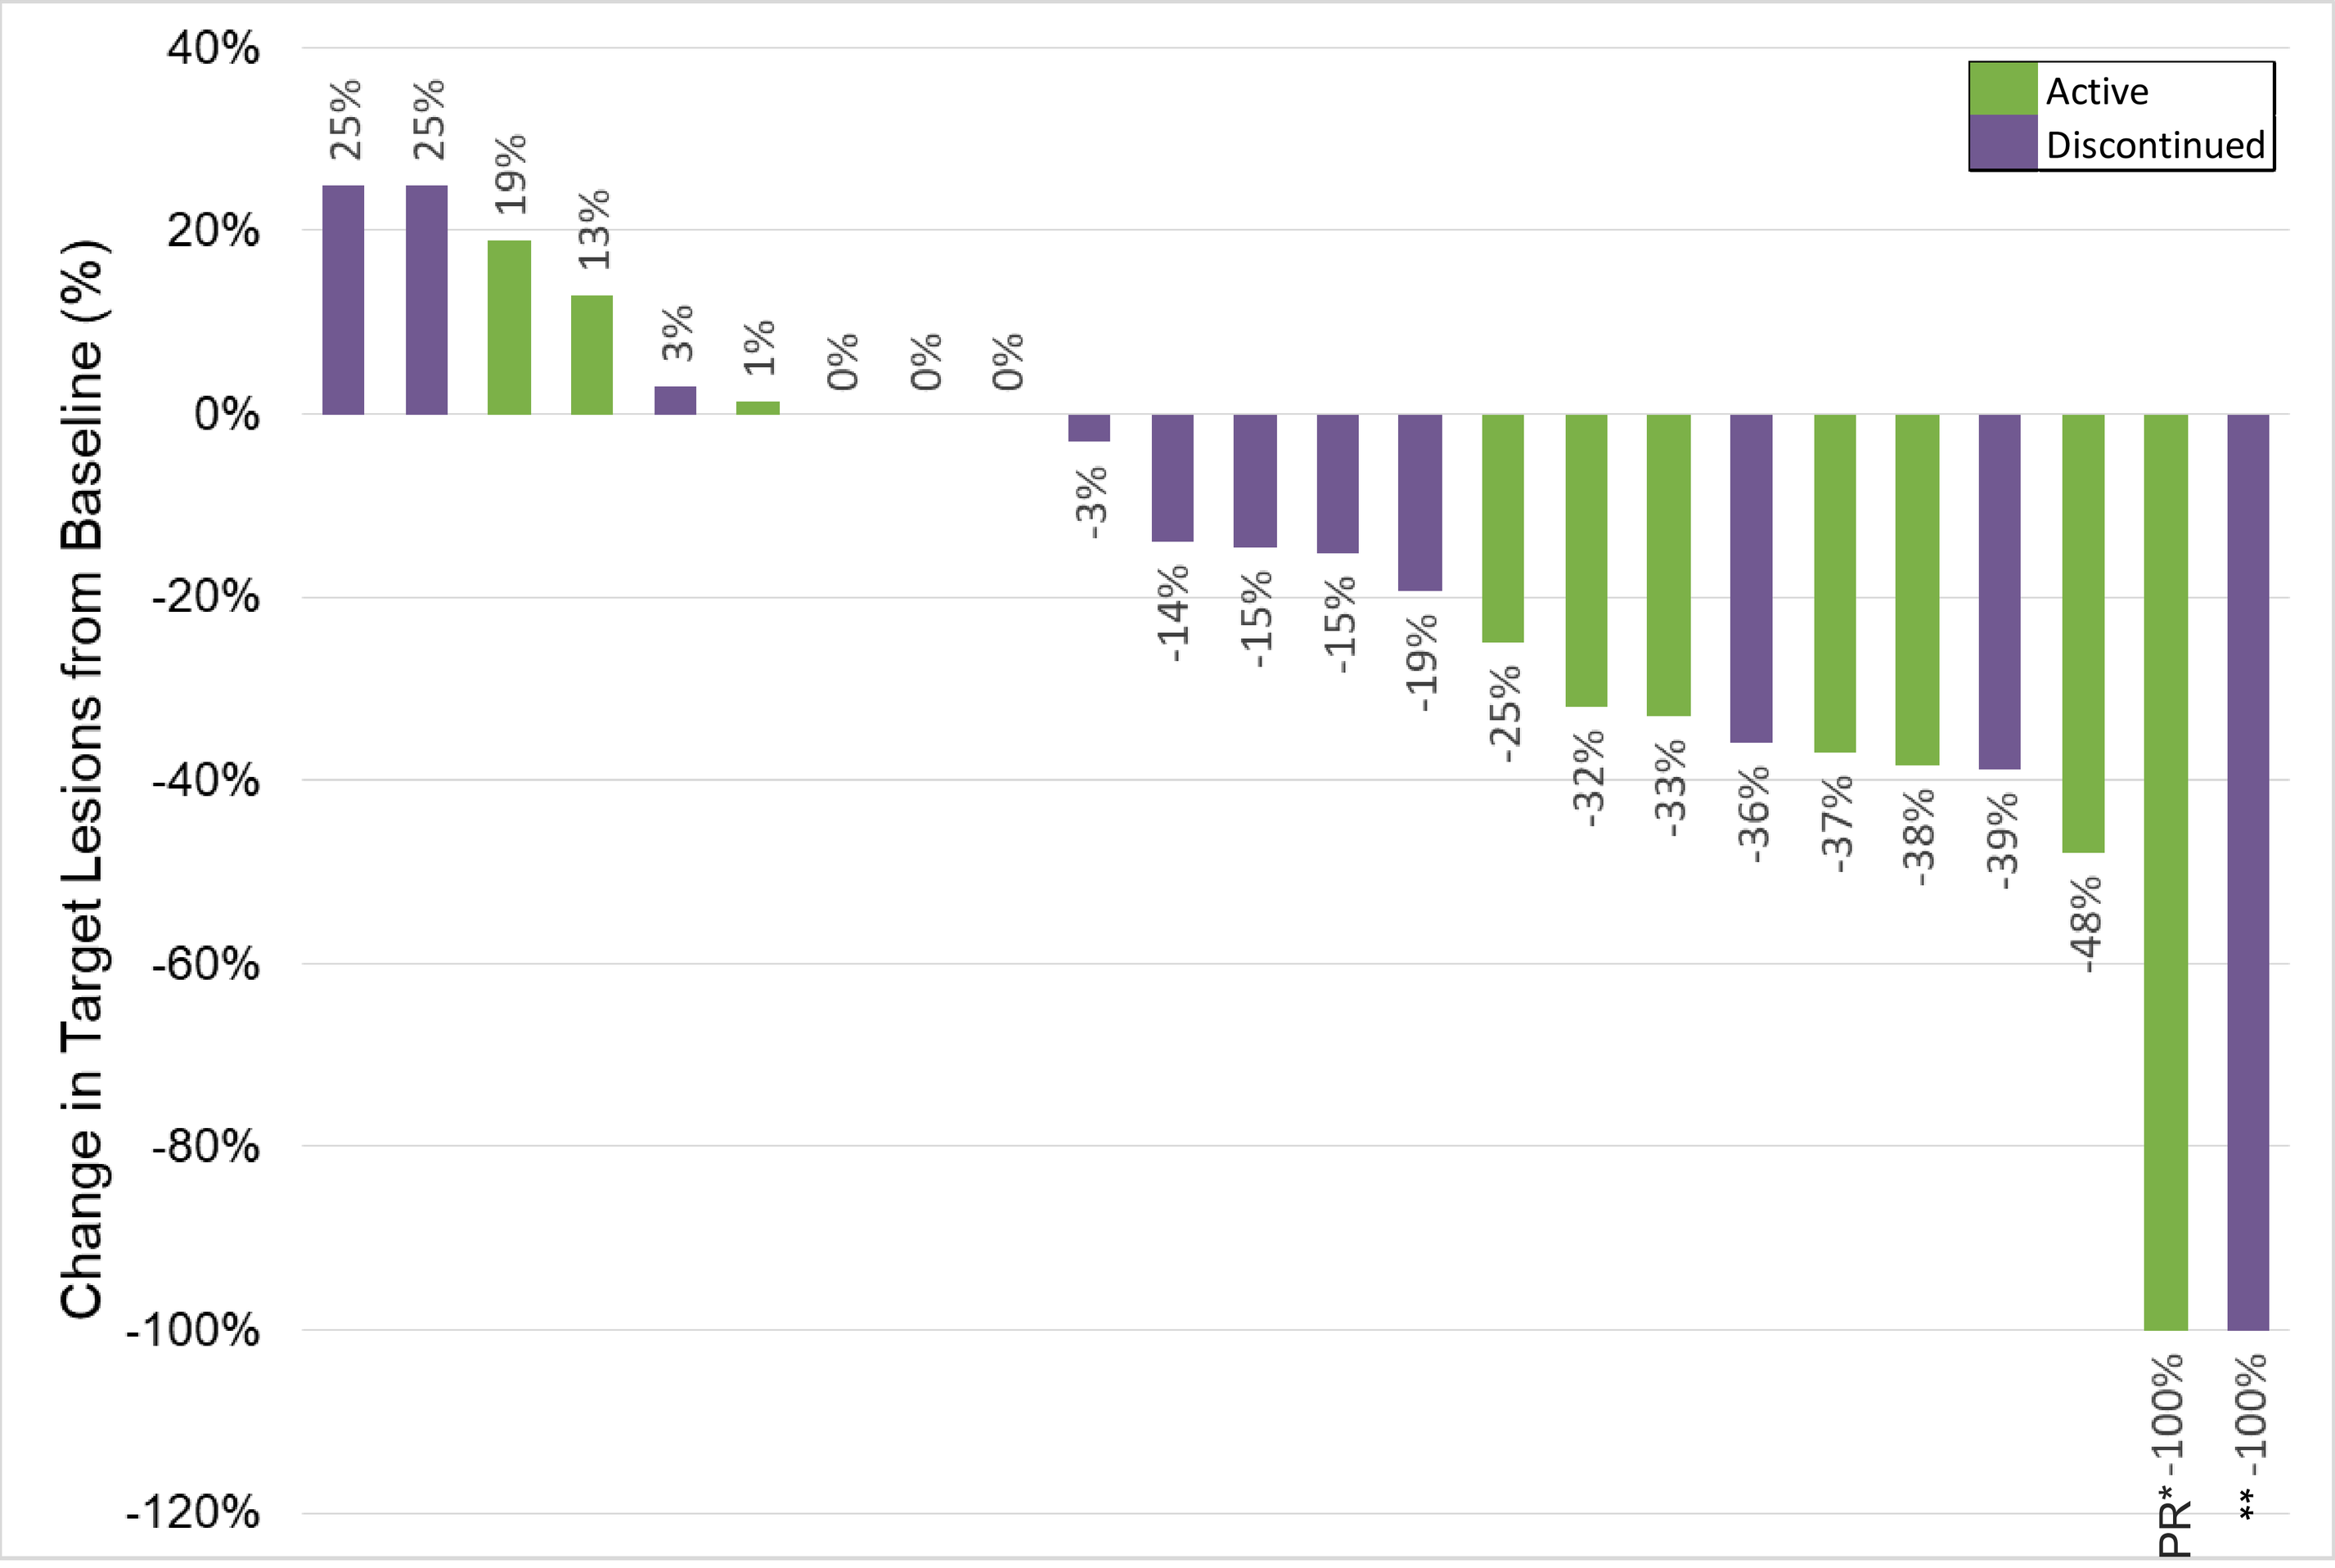
Change in Target Lesions from Baseline (%)
40%
20%
0%
-20%
-40%
-60%
-80%
-100%
-120%
25%
25%
19%
13%
3%
1%
0%
0%
0%
-3%
-14%
-15%
-15%
-19%
-25%
-32%
-33%
-36%
-37%
-38%
-39%
-48%
PR* -100%
** -100%
Active
Discontinued

*Small non-target lesions present
**Subject discontinued at Cycle 13 with a complete response (withdrew consent)
Note: Six subjects discontinued prior to the first on-study scan, 5 subjects are currently on treatment but have not completed the first on-study imaging assessment.

- As of 09 January 2019, in the response evaluable population (24 subjects) from the Phase 1 and Phase 2:
 - Overall response rate was 38%
 - 1 CR was observed after 6 cycles of therapy and confirmed after 8 cycles of therapy
 - 8 PRs were observed: 4 after 2 cycles of therapy and 4 after 4 cycles of therapy
 - Disease control rate was 92% at 8 weeks (1 Complete Response/ 8 Partial Responses/ 13 Stable Disease)
 - Most subjects (69%) with CA 19-9 results exhibited reductions after 1 cycle of therapy (-10% to -75%). One subject experienced a 46% increase but had a partial response after 4 cycles.

Pharmacokinetics

PK Parameter	nab-Pac		RX-3117	
	Cycle 1 Day 1	**Cycle 1 Day 15**	**Cycle 1 Day 1**	**Cycle 1 Day 15**
AUC (0-24) (hr×ng/mL)	4,765 [37.9] (17)	—†	9,268 [63.3] (15)	—†
AUC (0-6) (hr×ng/mL)	3,380 [45.7] (17)	2,842 [71.6] (14)	3,574 [48.5] (17)	2,432 [46.7] (14)
Cmax (ng/mL)	3,297 [64.0] (17)	2,402 [112.1] (14)	948 [45.5] (17)	628 [43.0] (14)
Tmax (hr)	0.50 (17) [0.00 – 0.50]	0.50 (14) [0.00 – 0.50]	2.00 (17) [1.00 – 4.00]	3.00 (14) [1.00 – 6.00]

*Geometric mean [geometric %CV] (N) except Tmax for which the median (N) [Range] are reported.
†A 24-hour sample was not collected after the dose on Day 15.

CA19-9 Percent Change from Baseline Over Time

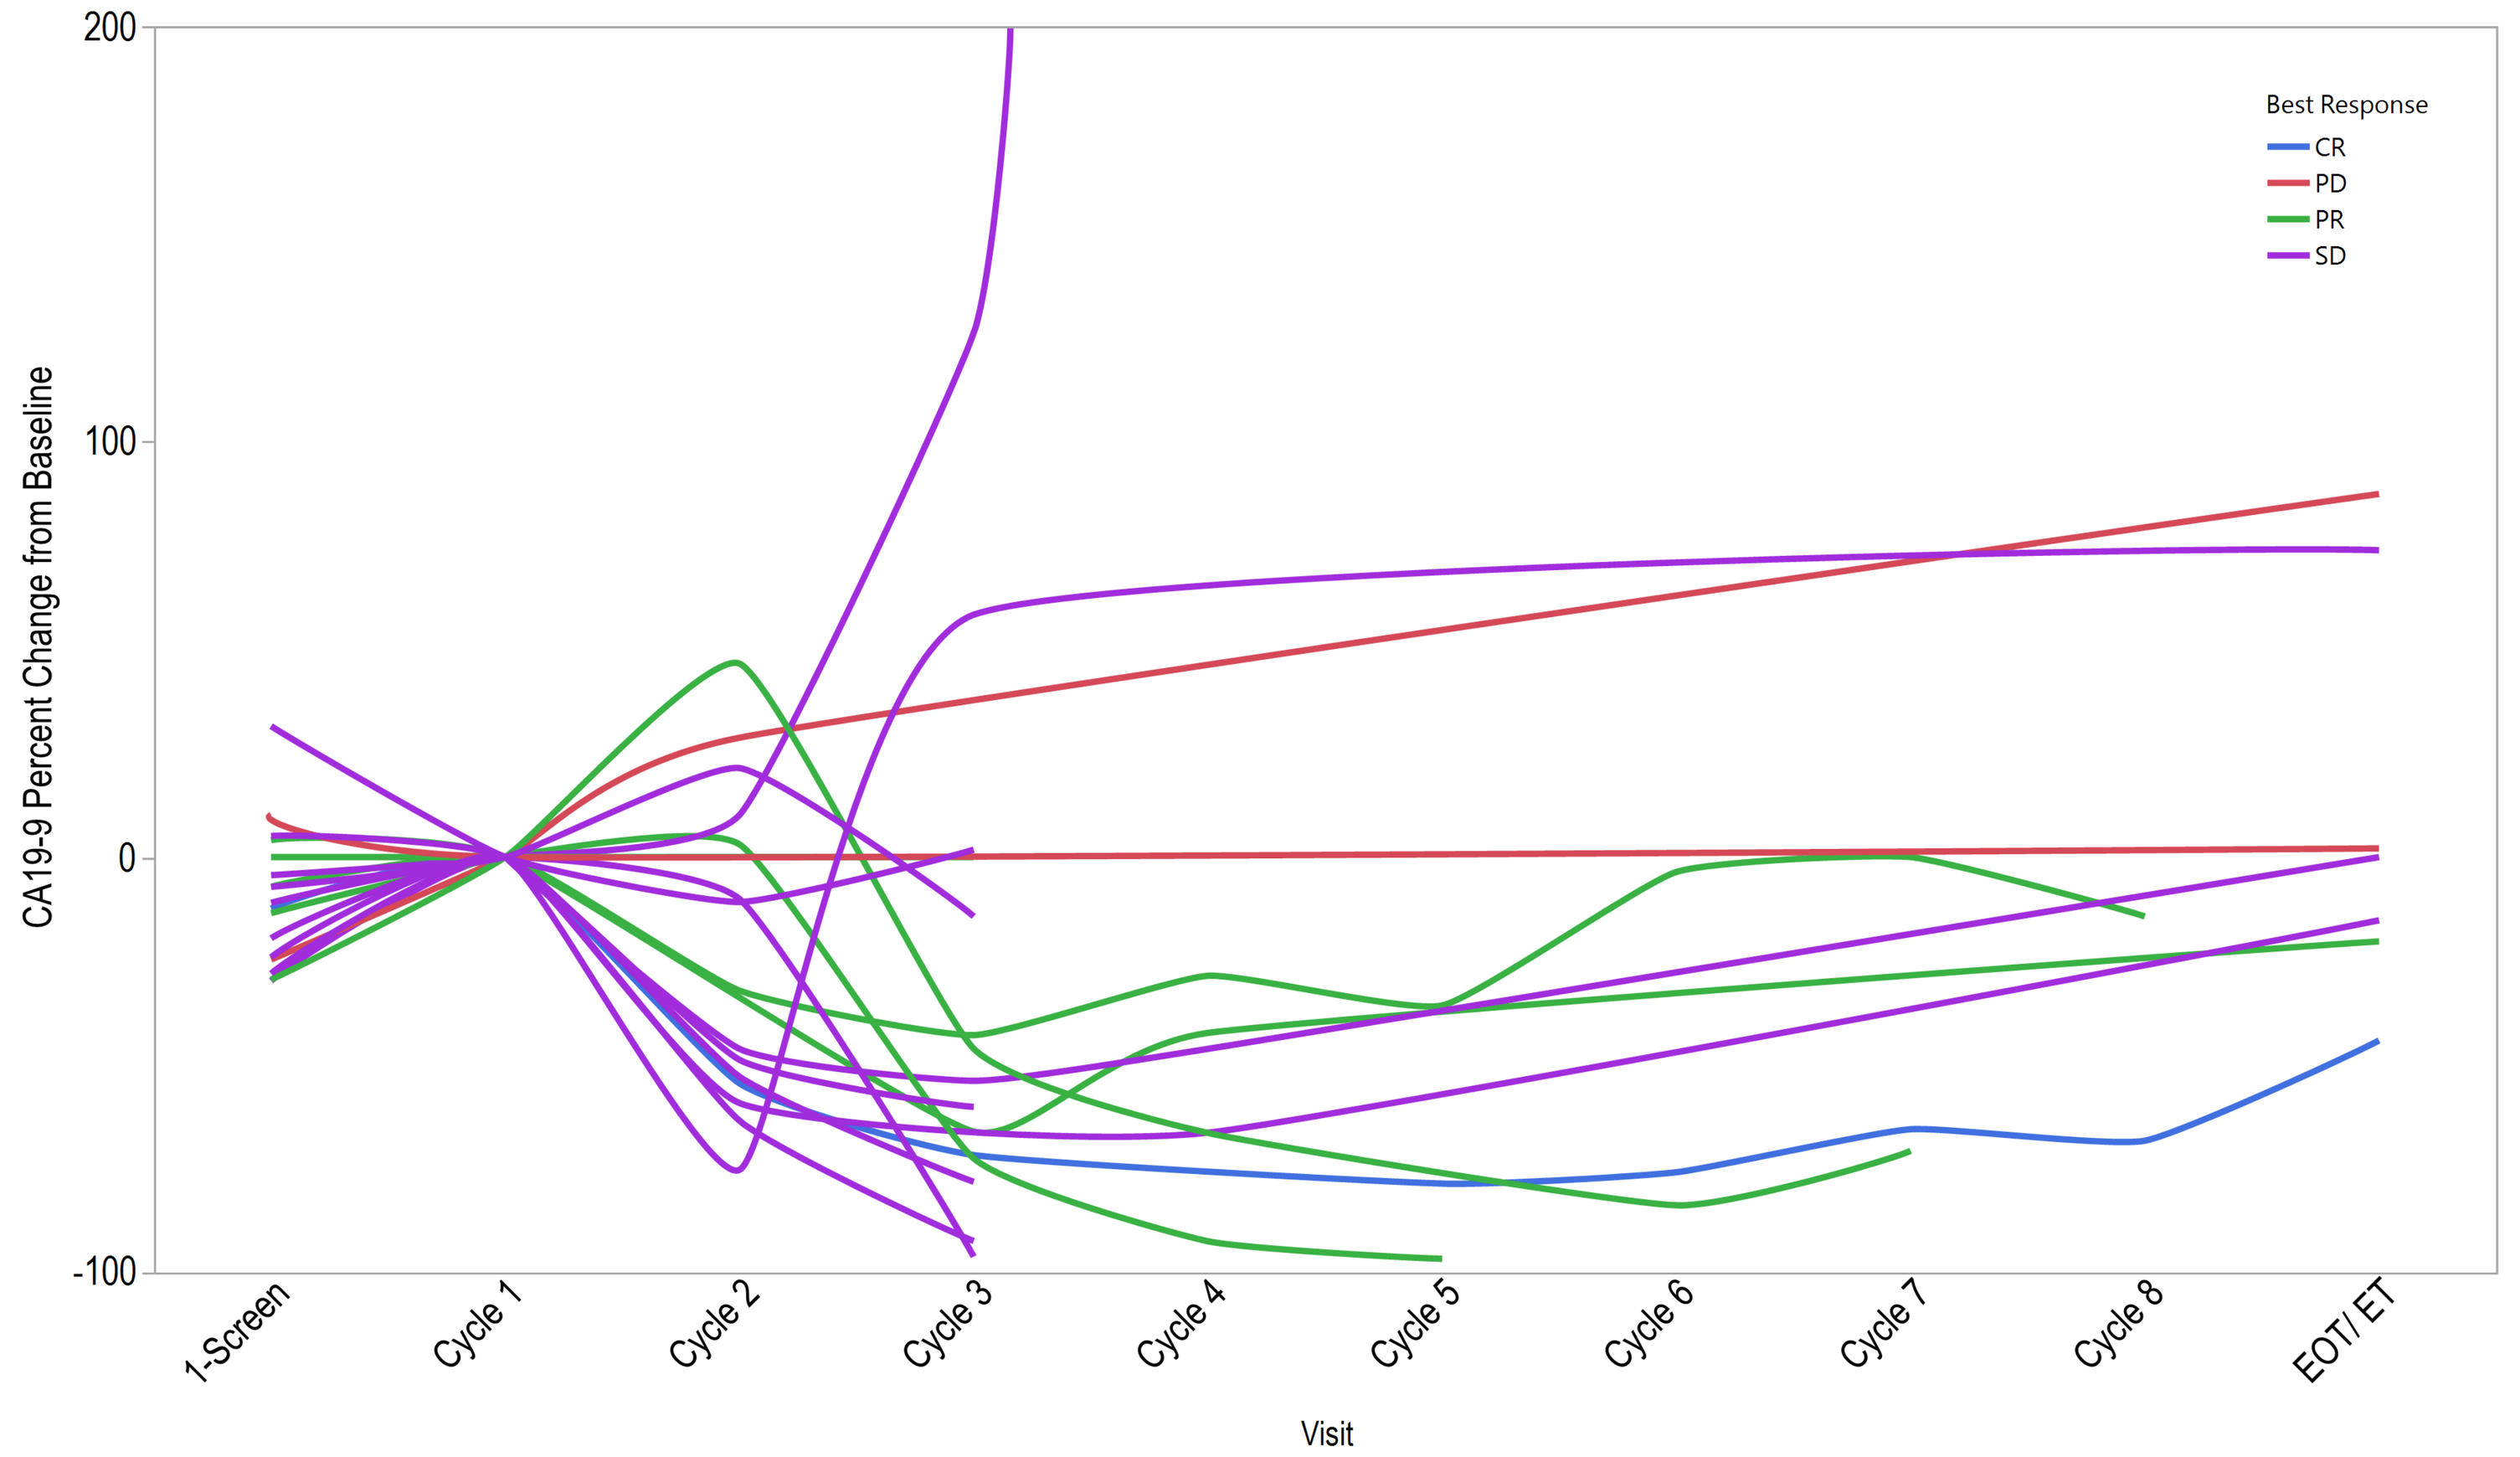
CA19-9 Percent Change from Baseline
200
100
0
-100
1-Screen
Cycle 1
Cycle 2
Cycle 3
Cycle 4
Cycle 5
Cycle 6
Cycle 7
Cycle 8
EOT/ ET
Visit
Best Response
CR
PD
PR
SD

Safety

- Most Treatment Emergent Adverse Events (TEAEs) considered related to RX-3117, nab-Pac, or the combination were mild to moderate (83%).
- The most common related Grade 1-2 TEAEs were diarrhea (53%), nausea (47%), fatigue (42%), rash (28%), alopecia (25%), anemia (19%), anorexia (19%), peripheral sensory neuropathy (19%), abdominal discomfort (17%), pyrexia (14%), constipation (11%), dehydration (11%), itching (11%), and vomiting (11%).
- Grade 3-4 TEAEs related to RX-3117, nab-pac, or the combination were neutropenia (29%), anemia (11%), diarrhea (8%), leucopenia (8%), hypokalemia (5%), and acute kidney injury, dehydration, general weakness, hypomagnesemia, mouth sores, nausea, neutropenic fever, peripheral sensory neuropathy, platelet count decreased, protein-calorie malnutrition, vomiting (1%).
- Some subjects received at least one dose reduction of nab-Pac (10/ 28%) or RX-3117 (2/ 5.6%). Dose reductions occurred most frequently in Cycles 1-3 (40%, 30%, 20% respectively).

Conclusions

- The combination of RX-3117 and nab-pac appears safe and well-tolerated when administered at the recommended Phase 2 dose
- Early responses were detected with an overall response rate of 38% in 24 subjects, and a disease control rate of 92% at 8 weeks.
- Pharmacokinetic results indicate that RX-3117 and nab-Pac do not appear to interfere with the exposure or clearance of either drug.
- The study continues to enroll subjects with metastatic pancreatic cancer in Stage 2.

Author Disclosures: Ely Benaim, MD, Callie Heaton, and Christine Peterson, PhD – Rexahn Pharmaceuticals
For further information about RX-3117 and Rexahn Pharmaceuticals
please contact Dr. Ely Benaim: benaime@rexahn.com, (240) 268-5300 x 304

INSERT QR CODE HERE