Safety and Efficacy of Lenabasum (JBT-101) in an Open-Label Extension of a Phase 2 Study in Diffuse Cutaneous Systemic Sclerosis Subjects

R. F. Spiera[1], L. K. Hummers[2], L. Chung[3], T. M. Frech[4], R. T. Domsic[5], V. Hsu[6], D. E. Furst[7], J. K. Gordon[1], M. D. Mayes[8], R. W. Simms[9], E. Lee[10], N. Dgetluck[10], S. Constantine[10], B. White[10]

[1]Hospital for Special Surgery, New York, NY. [2]Johns Hopkins University School of Medicine, Baltimore, MD. [3]Stanford University School of Medicine, Palo Alto, CA. [4]University of Utah, Salt Lake City, UT. [5]University of Pittsburgh, Pittsburgh, PA.
[6]Rutgers Robert Wood Johnson Medical School, New Brunswick, NJ. [7]Pacific Arthritis Associates, Los Angeles, CA. [8]University of Texas McGovern Medical School, Houston, TX. [9]Boston University School of Medicine, Boston, MA. [10]Corbus Pharmaceuticals, Inc., Norwood, MA

Poster # 1715

ABSTRACT

Background/Purpose: Lenabasum is a synthetic, non-immunosuppressive, selective cannabinoid receptor type 2 agonist that activates resolution of innate immune responses. Lenabasum had acceptable safety and tolerability, and improved multiple efficacy outcomes in the double-blinded, randomized, placebo-controlled Part A of Phase 2 trial JBT101-SSc-001 (NCT02465437) in diffuse cutaneous SSc (dcSSc) subjects.
Objective: To provide long-term open-label safety and efficacy data in dcSSc subjects in study JBT101-SSc-001.
Methods: Subjects who completed Part A were eligible to receive oral lenabasum 20 mg BID in an open-label extension (OLE) that assessed safety and efficacy at 4 weeks, then every 8 weeks.
Results: 36/38 (95%) eligible subjects enrolled in the OLE, with mean interval of 134 (range 33-392) days or 19.1 weeks from end of dosing in Part A to start of OLE when subjects received only standard-of care drugs. 34/36 (94%) subjects were on stable doses of immunosuppressive drugs. At the time of data cut-off, 5/36 (13.9%) subjects had discontinued the OLE for reasons all unrelated to lenabasum: difficulty coming for study visits (n = 2), fatigue, inflamed tendons, and high dose steroid-induced scleroderma renal crisis. Of the remaining 31 subjects, 27 (87.1%) had already completed ≥ 1 year of dosing in OLE. Adverse events (AEs, n = 180) occurred in 33/36 (91.7%) subjects, with 7/36 (19.4%) subjects having ≥ 1 AE related to lenabasum. No subject had a serious or severe AE related to lenabasum. Three serious AEs occurred: renal crisis, thumb fracture, and digital ulcer. AEs that occurred in ≥ 10% of subjects (n, % of subjects) were: upper respiratory tract infection (8, 22.2%), skin ulcer, arthralgias, urinary tract infection (5, 13.9% each), and diarrhea (4, 11.1%). Mild dizziness occurred in 3 (8.3%) subjects.
Improvement was seen in multiple physician- and patient-reported efficacy outcomes compared to study start and start of OLE, including Combined Response Index in diffuse cutaneous Systemic Sclerosis (CRISS), mRSS, HAQ-DI, Physician Global Assessment, skin symptoms, itch, and multiple PROMIS-29 domains. FVC % predicted was relatively stable. Compared to baseline at study start, the CRISS median score was 92% (23%, 100% IQR) at Week 52 and mRSS declined by mean (SD) = 9.4 (8.43) and 41.3% (32.7%) from baseline, with 35% of subjects newly achieving a low mRSS ≤ 10.
Conclusion: In OLE of Phase 2 trial JBT101-SSc-001, lenabasum continues to have acceptable safety and tolerability in dcSSc with no severe or serious AEs or study discontinuations related to lenabasum. Only about 1 in 5 subjects had an AE related to lenabasum over 1-year OLE dosing. ACR CRISS score, mRSS, Physician Global Assessment, and multiple patient-reported outcomes show continued improvement, although background therapy, potential for spontaneous improvement, and open-label dosing limit what can be definitely attributed to lenabasum.

RATIONALE, STUDY DESIGN, AND SUBJECT CHARACTERISTICS

- Systemic sclerosis (SSc) is a serious systemic autoimmune disease characterized in part by chronic activation of innate immune responses accompanied by fibrosis.
- There is a major unmet need for treatments that are more effective and safe enough to treat a broad spectrum of individuals with diffuse cutaneous SSc.
- Lenabasum (JBT-101) is an oral selective cannabinoid receptor type 2 (CB2) agonist that resolves tissue inflammation and fibrotic processes without immunosuppression.
- Lenabasum showed promising safety and efficacy in a Phase 2 trial, JBT101-SSc-001. In JBT101-SSc-001, lenabasum was tested in a double-blind, placebo-controlled (DBPC) 4 month study, which was followed by a period off study drug (mean 5 months duration), and then followed by ongoing open-label dosing.
 - ✓ **Subjects**: Adults with dcSSc. Disease duration ≤ 3 years or > 3 and ≤ 6 years if mRSS ≥ 16 or high CRP or IL-6. Stable doses of concomitant medicines allowed, including immunosuppressive drugs
 - ✓ **DBPC Dosing**: Month 1: Lenabasum 5 mg QD, 20 mg QD 20 mg BID, or placebo. Months 2-3: 20 mg BID or placebo. Month 4: Safety and efficacy assessments continued
 - ✓ **Off Study Drug**: Background immunosuppressant medications continued, but no study drug was given.
 - ✓ **OLE Dosing**: Lenabasum 20 mg BID for subjects who completed DBPC dosing with lenabasum or placebo

DBPC 4 months → Off study drug mean 5 months → **OLE** 18 months

Baseline demographics and disease characteristics

Characteristic	Double-blinded Dosing		Open-label Dosing
	Lenabasum n = 27	Placebo n = 15	Lenabasum N = 36
Female, %	85%	60%	75%
Age, mean (SD)	49 (10.4)	47 (11.1)	49 (11.2)
Caucasian, %	82%	80%	83%
Disease duration, months, mean (SD)	34 (16.6)	34 (18.0)	43 (17.5)
Concomitant immunosuppressive drugs, %	93%	87%	92%
Modified Rodnan skin score, mean (SD)	24 (10.4)	26 (11.2)	20 (10.9)

Safety and efficacy data after 18 months OLE dosing are being presented.

ADVERSE EVENTS DURING OPEN-LABEL DOSING

- There were no serious adverse events (AEs) related to lenabasum and no deaths in the study to date
- The only AE related to lenabasum that occurred in more than 1 subject during the OLE was dizziness, which occurred in 2 (6%) subjects to date
- 34 (94%) subjects had at least 1 AE, with 213 total AEs in the 36 subjects during the OLE to date
- By maximum severity, 5 (14%) of subjects had mild AEs, 25 (69%) had moderate AEs, 3 (8%) had severe AEs, and 1(3%) had a life-threatening AE of renal crisis caused by high-dose steroids
- By maximum relatedness, 27 (75%) of subjects had AEs unrelated to lenabasum and 7 (19%) had AEs related to lenabasum. The rate of AEs related to lenabasum decreased with time
- AEs (n, % 36 subjects) occurring in ≥ 10% of subjects during the OLE to date (18 months) were: upper respiratory tract infection 10 (28%); skin ulcer 6 (17%); urinary tract infection and arthralgia 5 (14%) each; and diarrhea 4 (11%)
- 83% of subjects who entered the OLE remained in the study at 18 months

EFFECTS ON OVERALL DISEASE

Effects of lenabasum on overall disease were evaluated with 3 outcomes:

- ACR CRISS score, a weighted composite of change from baseline in modified Rodnan skin score (mRSS), patient global assessment of health (PtGA), physician global assessment of health (MDGA), health assessment questionnaire disability index (HAQ-DI, and forced vital capacity (FVC), % predicted
- PtGA on a 10-cm visual analogue scale (VAS)
- MDGA, on a 10-cm VAS

ACR CRISS Score

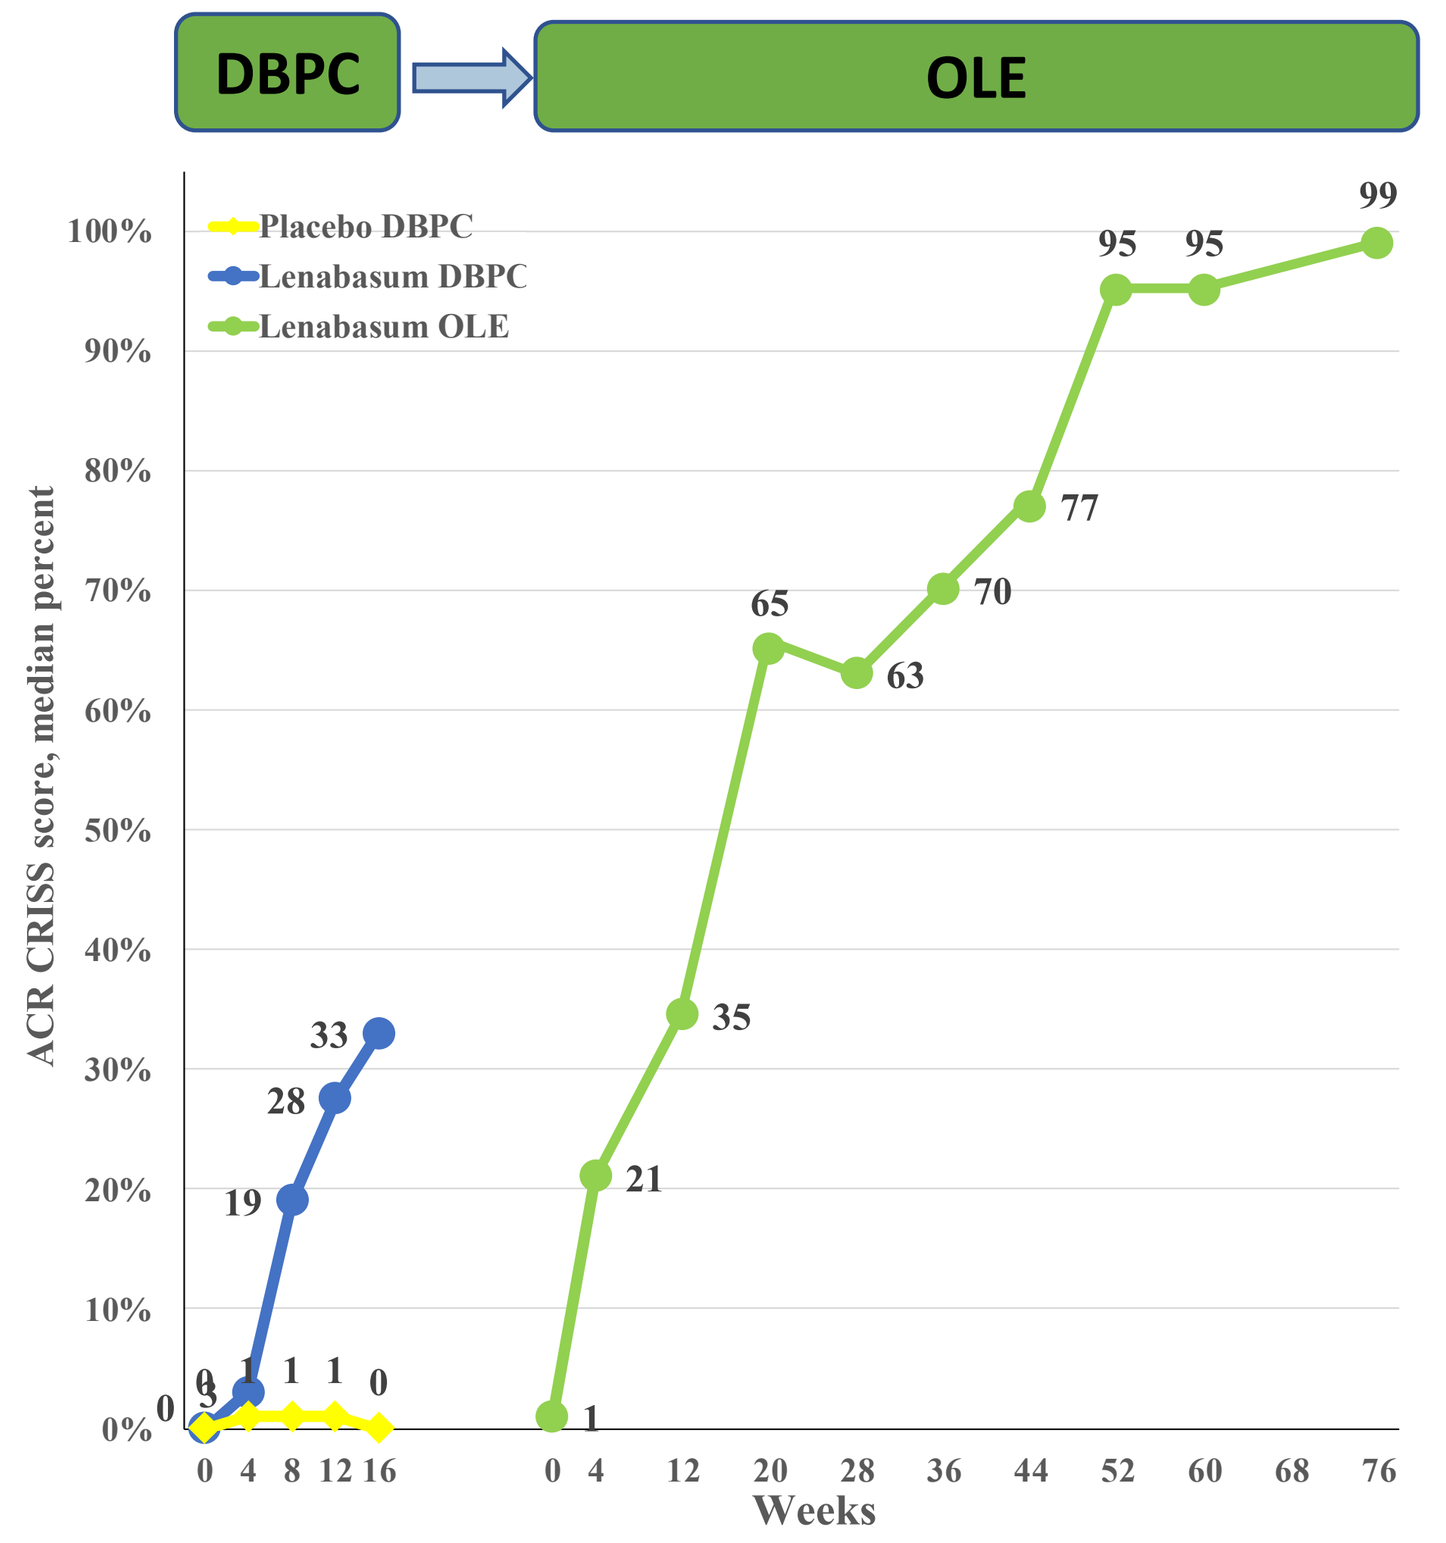

DBPC
OLE
Placebo DBPC
Lenabasum DBPC
Lenabasum OLE
ACR CRISS score, median percent
Weeks

Distribution of ACR CRISS Scores by Individual Subject, OLE Month 18

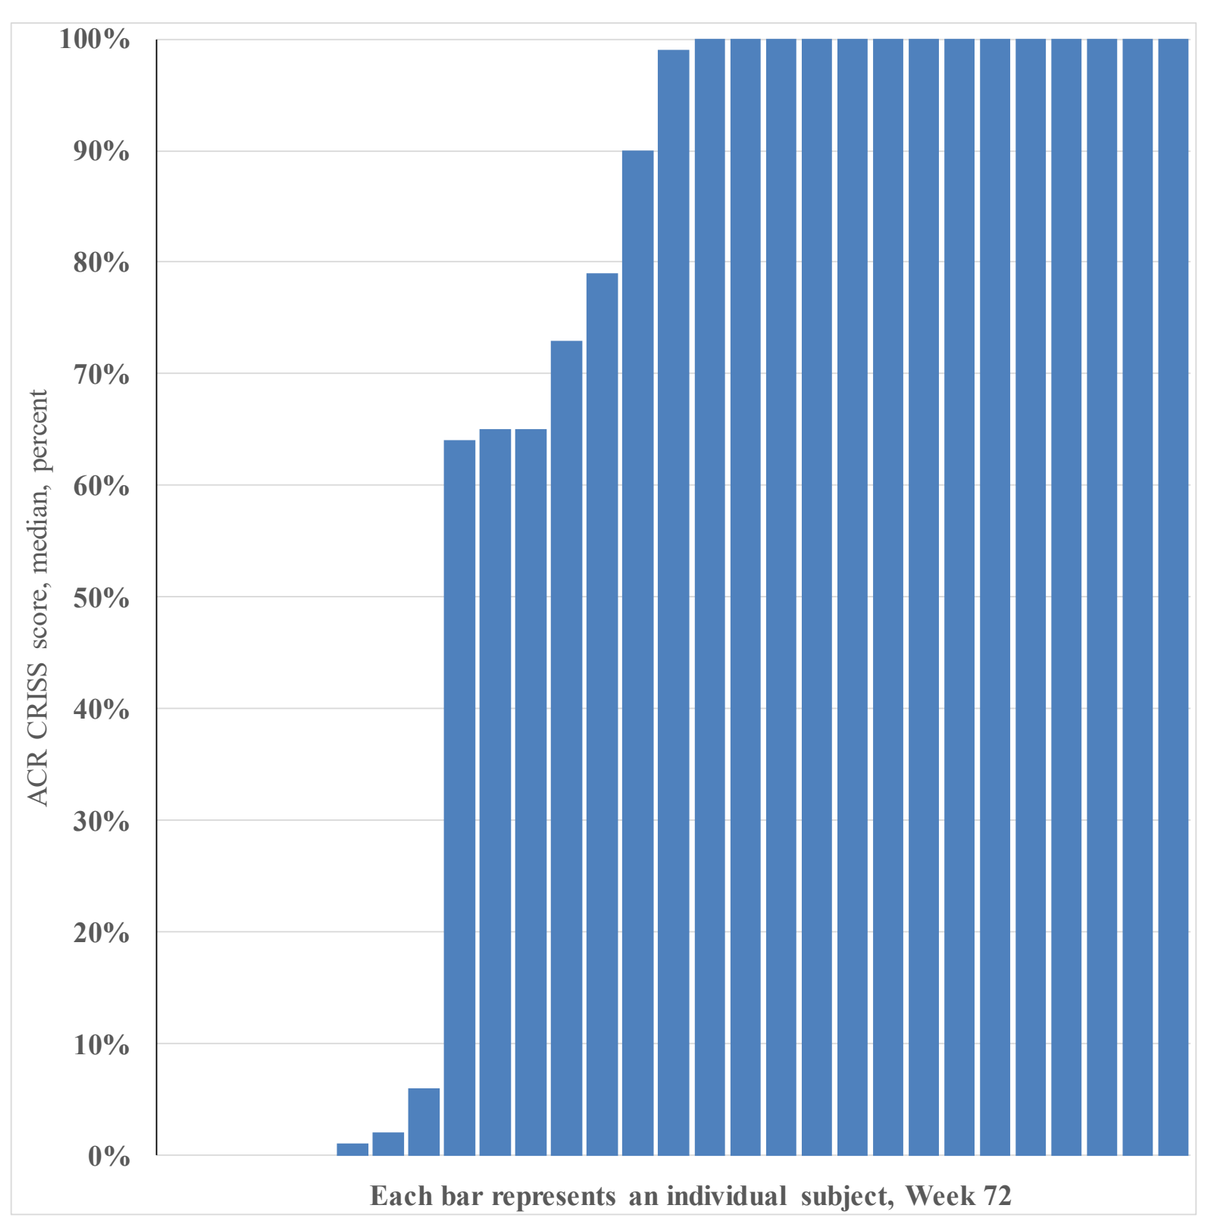

ACR CRISS score, median, percent
Each bar represents an individual subject, Week 72

- ACR CRISS responses increased during the OLE and were durable, with median ACR CRISS score ≥ 95% from 12-18 months dosing
 - ✓ Median ACR CRISS reached 99% (6%, 100% IQR) at 18 months, stable from 95% (45%, 100% IQR) at 12 months
- About 3 in 4 subjects achieved an ACR CRISS score ≥ 60% at 18 months
- About 1 in 2 subjects achieved an ACR CRISS score = 100% at 18 months

Patient Global Assessment / Physician Global Assessment

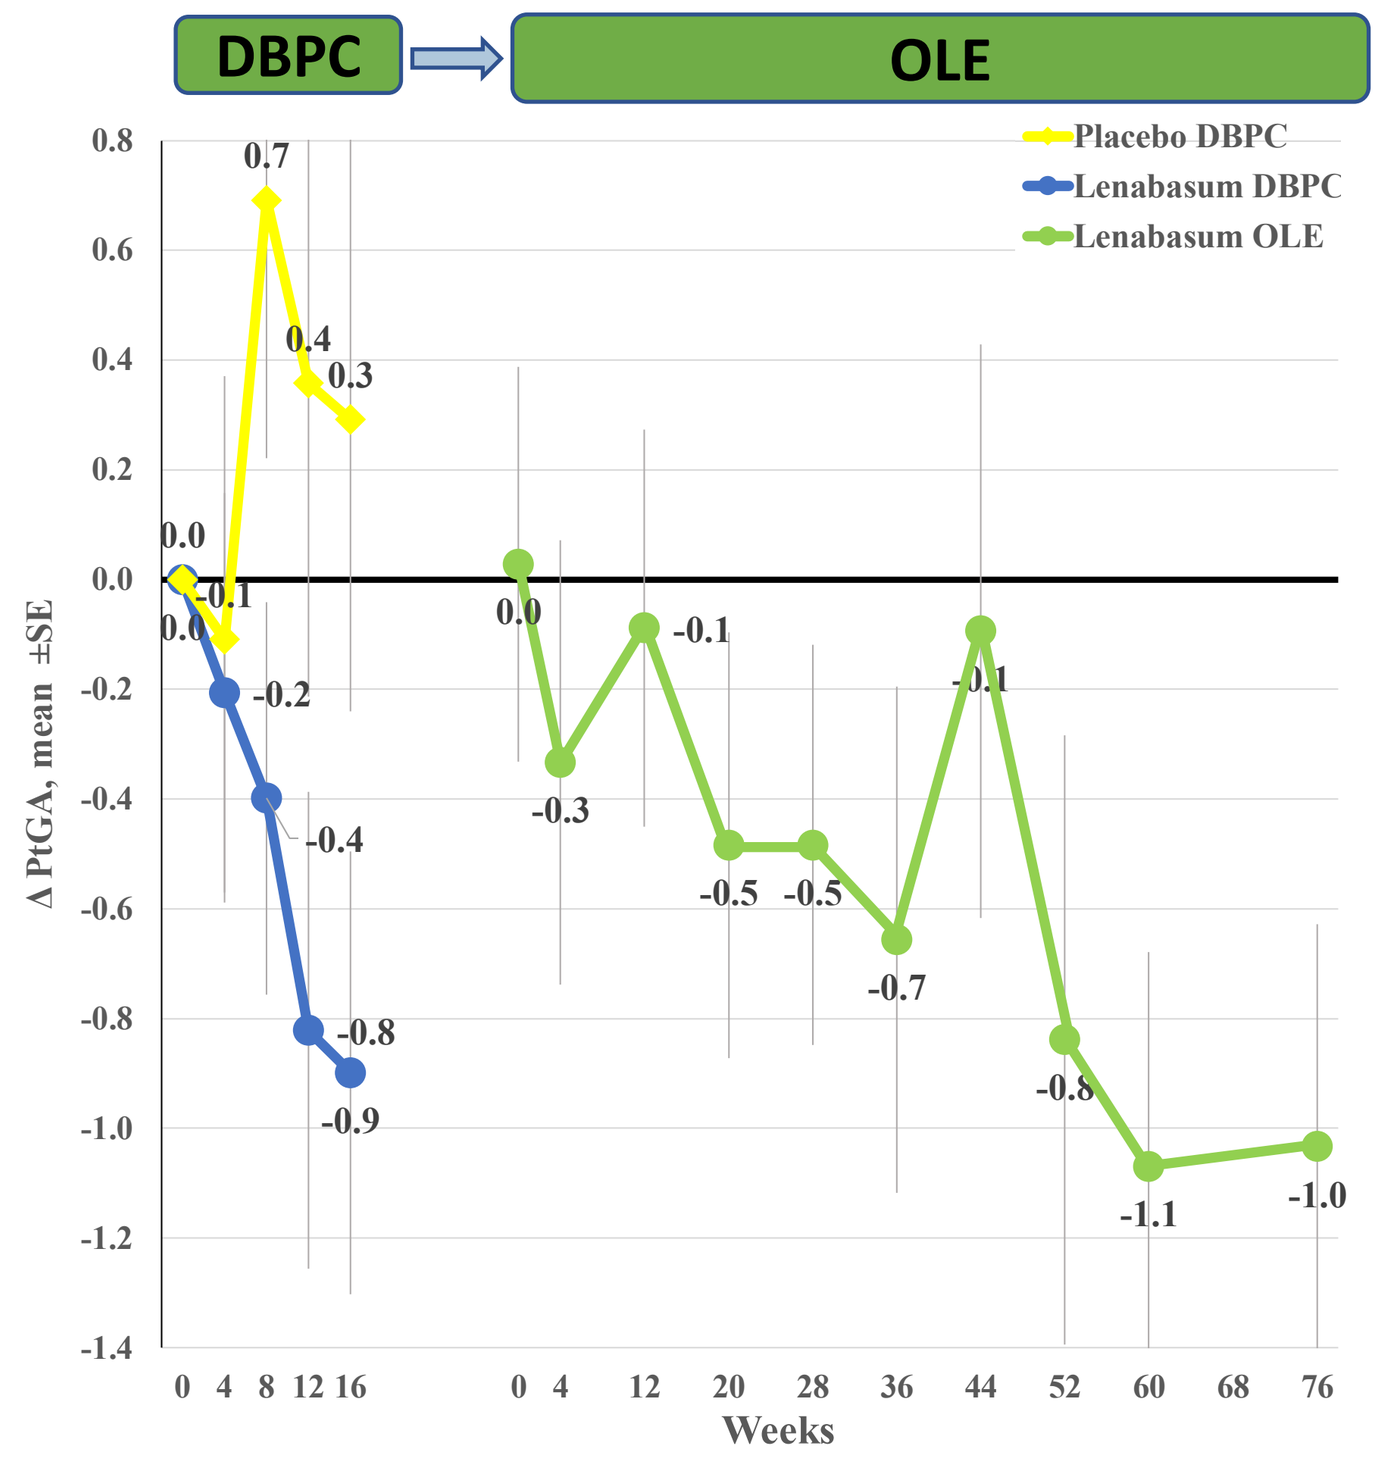

DBPC
OLE
Placebo DBPC
Lenabasum DBPC
Lenabasum OLE
Δ PtGA, mean ±SE
Weeks

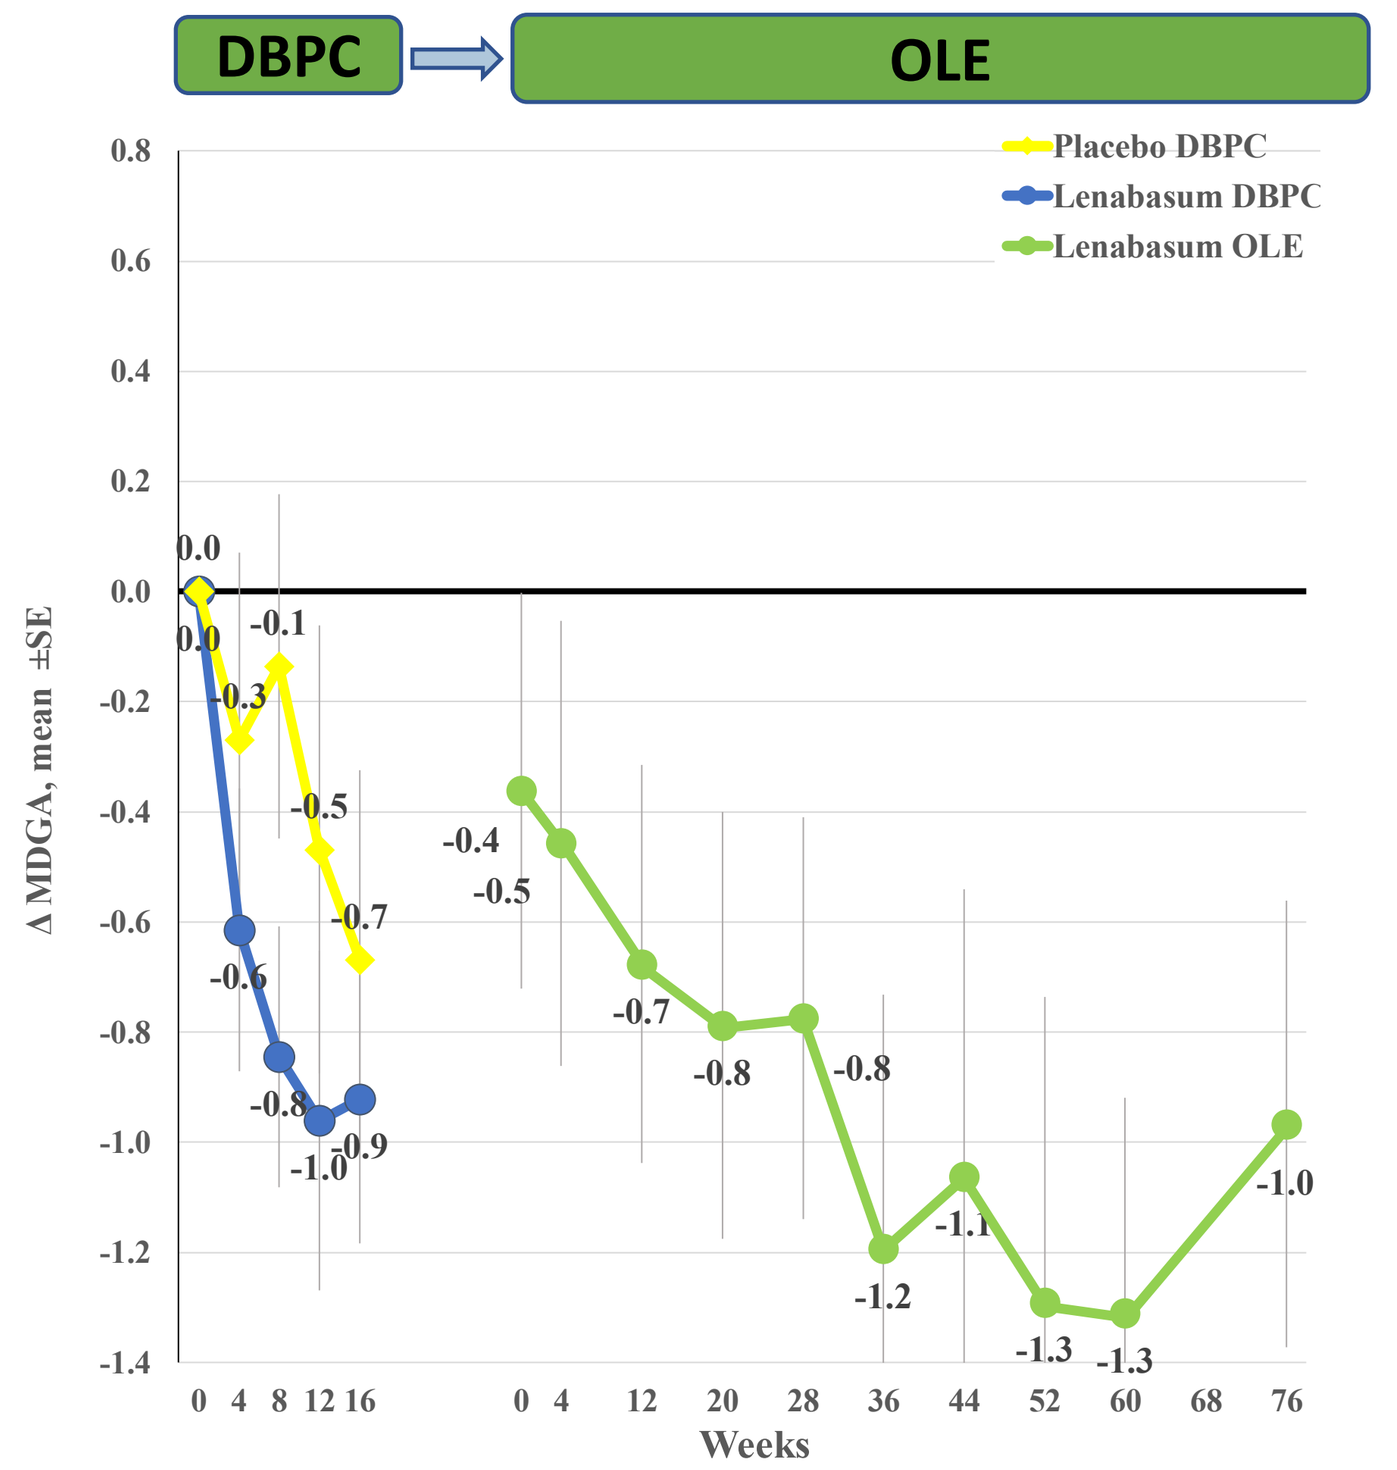

DBPC
OLE
Placebo DBPC
Lenabasum DBPC
Lenabasum OLE
Δ MDGA, mean ±SE
Weeks

- As assessed with PtGA and MDGA, overall disease improved during the OLE, reaching a mean (SD) of -1.0 in PtGA and -1.0 in MDGA at 18 months
- Improvements were durable, with general stability in PtGA and MDGA from Months 12-18

EFFECTS ON SKIN

Effects of lenabasum on skin involvement were evaluated with 3 outcomes:

- mRSS, change from baseline
- SSPRO-18 – a patient-reported outcome of impact on skin on quality of life
- 5-D Itch score – questionnaire that assess severity of itch in skin diseases

Modified Rodnan Skin Score

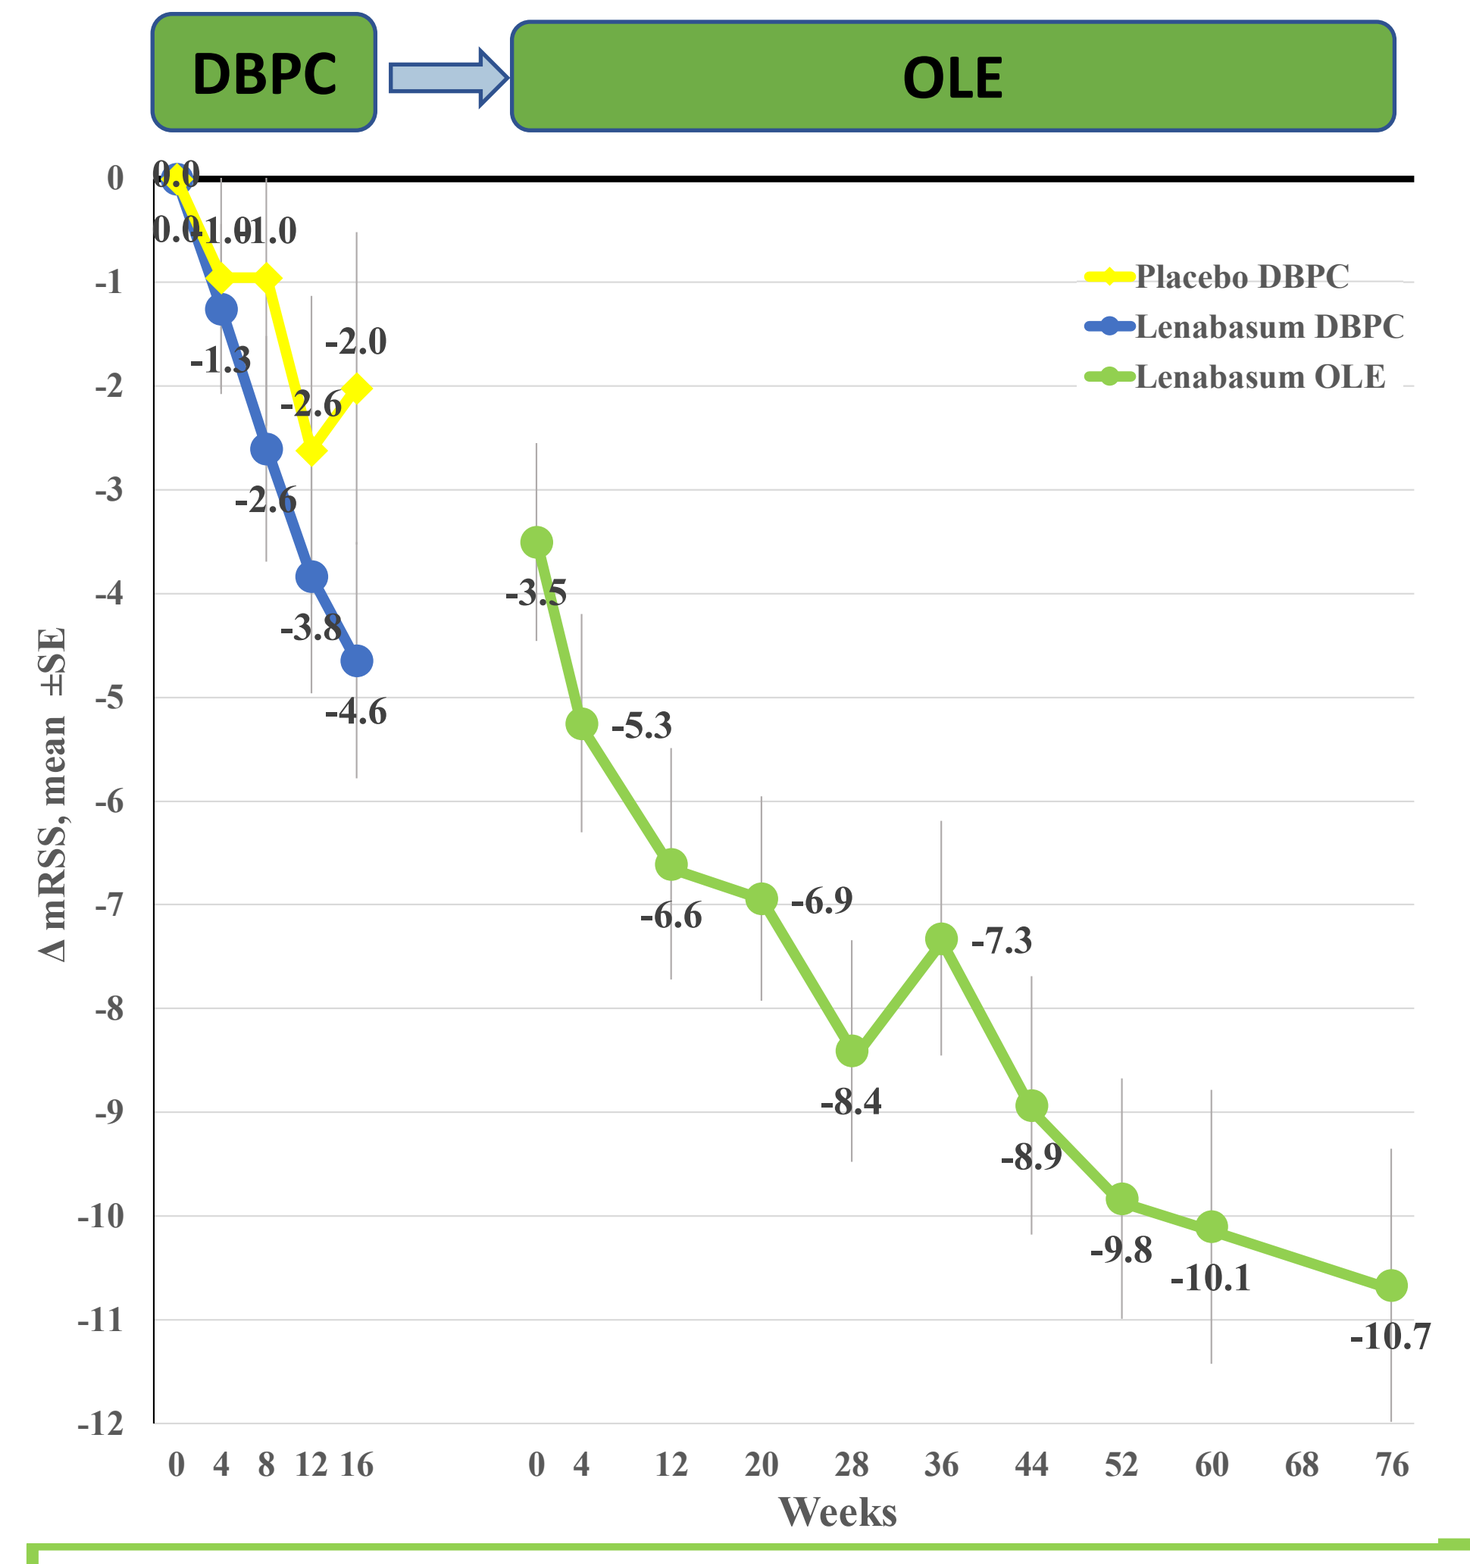

DBPC
OLE
Placebo DBPC
Lenabasum DBPC
Lenabasum OLE
Δ mRSS, mean ±SE
Weeks

Distribution of change from Baseline by Individual Subject, OLE Month 18

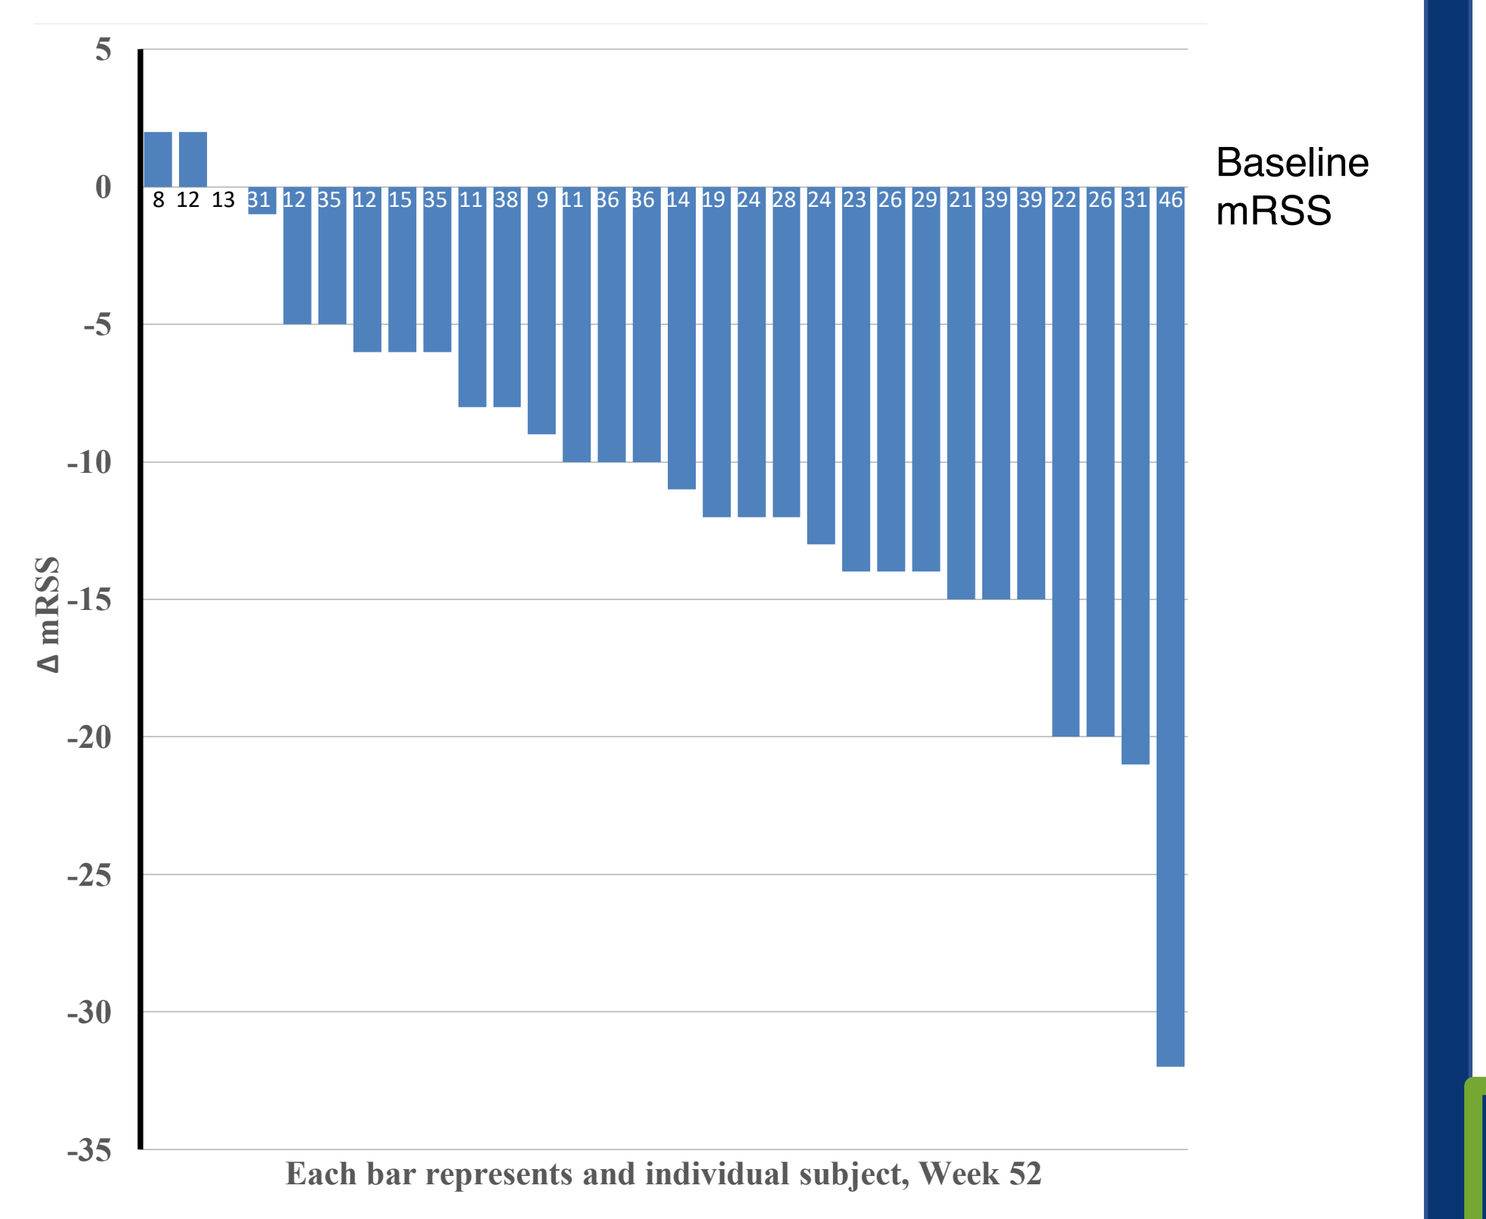

Baseline mRSS
Δ mRSS
Each bar represents and individual subject, Week 52

- mRSS scores improved during the OLE and were durable, with mean mRSS score about -10 points from months 12-18
- About half (47%) of subjects achieved a low absolute mRSS of ≤ 10 points
- 60% of subjects achieved a change from Baseline in mRSS score of ≤ -10 points at 18 months
- 87% achieved a minimal important difference in change from Baseline in mRSS score of ≤ -5 points at 18 months

SSPRO-18 Score / 5-D Itch score

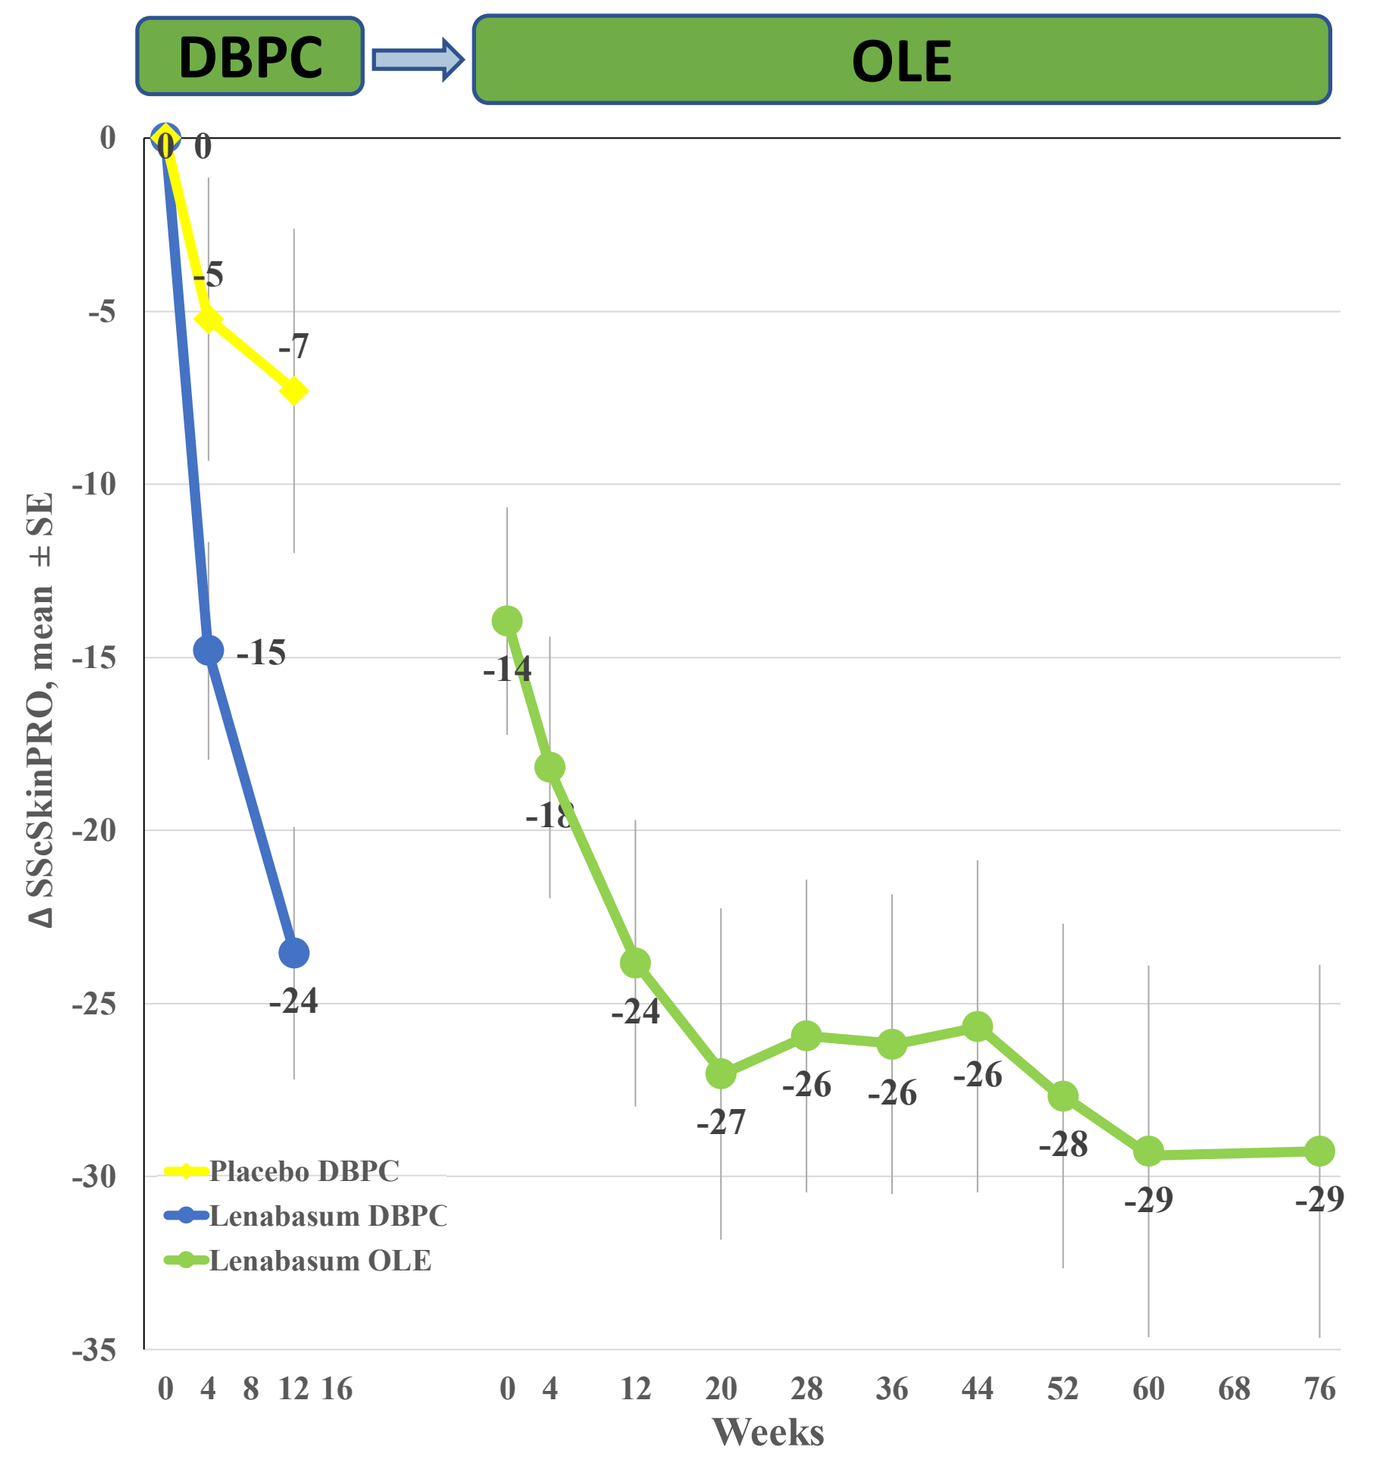

DBPC
OLE
Placebo DBPC
Lenabasum DBPC
Lenabasum OLE
Δ SScSkinPRO, mean ± SE
Weeks

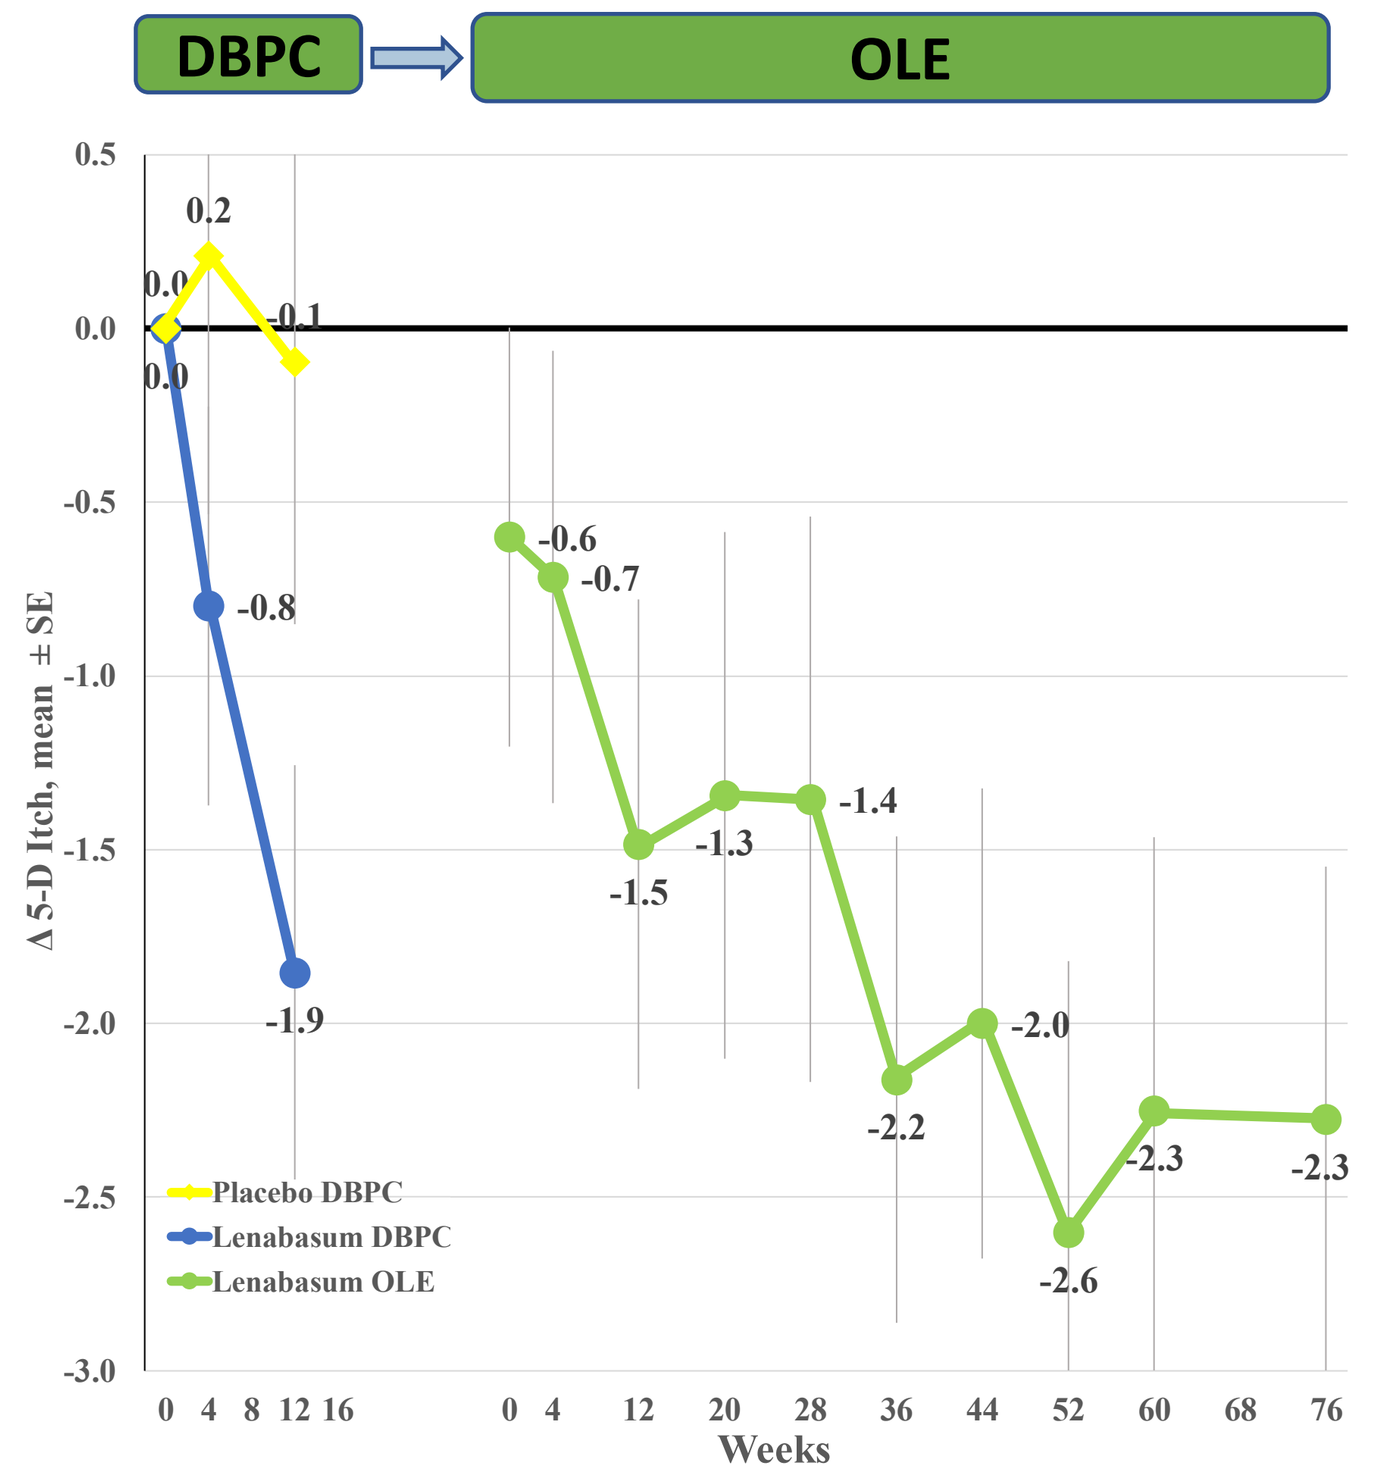

DBPC
OLE
Placebo DBPC
Lenabasum DBPC
Lenabasum OLE
Δ 5-D Itch, mean ± SE
Weeks

- As assessed with two patient-reported outcomes, SSPRO-18 and 5-D Itch score, skin involvement improved steadily during the OLE
- Improvements were durable, with general stability in PtGA and MDGA from Months 12-18

EFFECTS ON FUNCTION

Effects of lenabasum patient were evaluated with 3 outcomes:

- HAQ-DI, a measure of patient-reported disability
- PROMIS-29 Physical function domain
- PROMIS-29 Social role domain, a measure of social functioning

HAQ-DI / PROMIS-29 Physical Function / PROMIS-29 Social Role

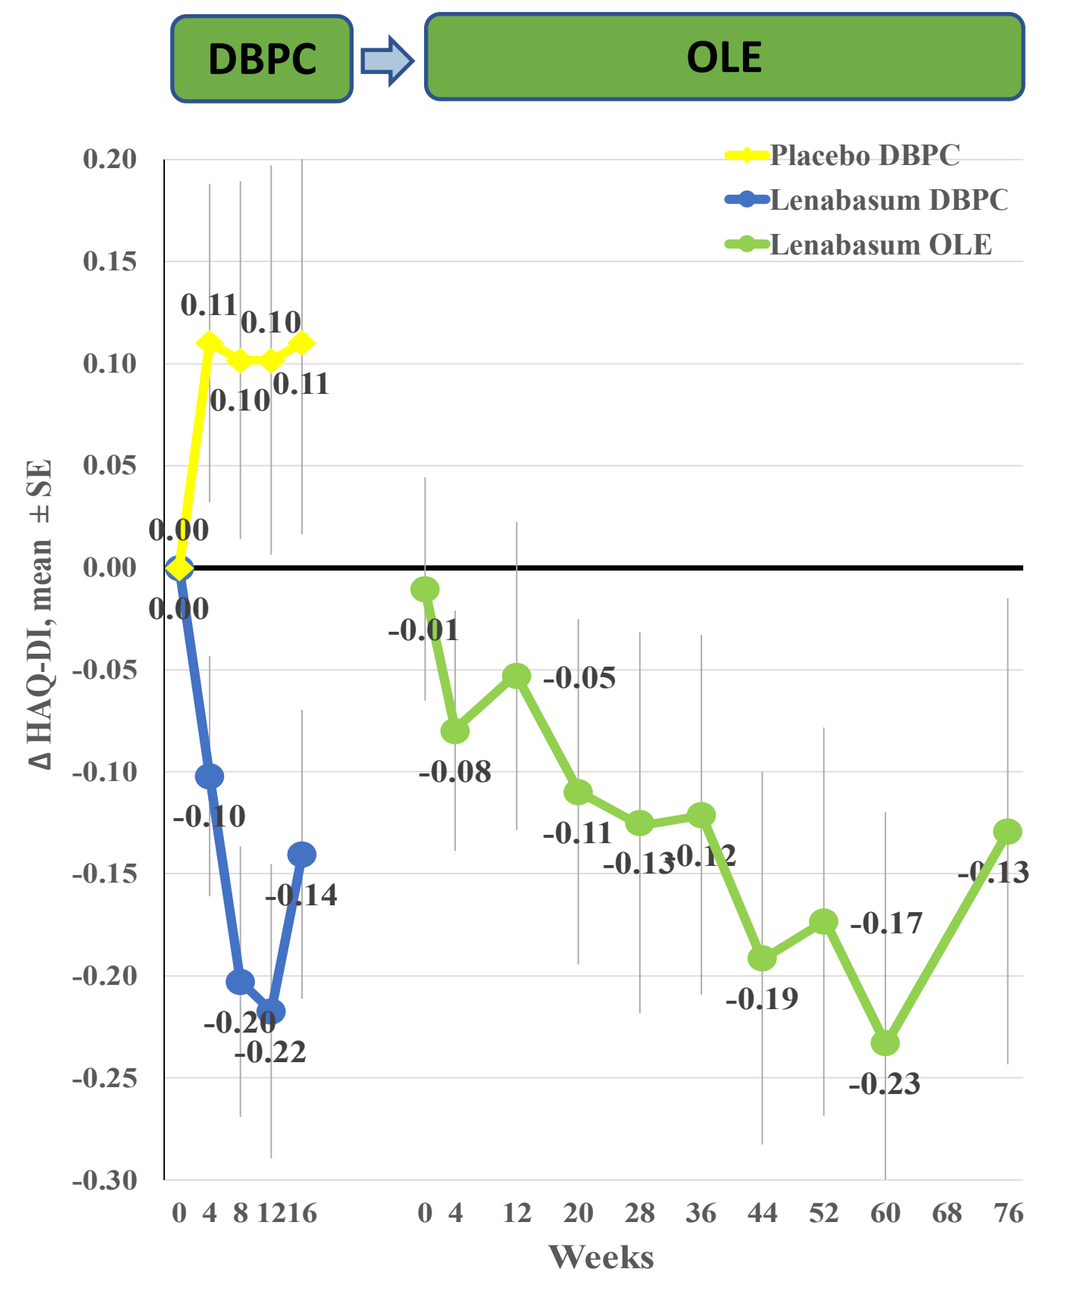

DBPC
OLE
Placebo DBPC
Lenabasum DBPC
Lenabasum OLE
Δ HAQ-DI, mean ± SE
Weeks

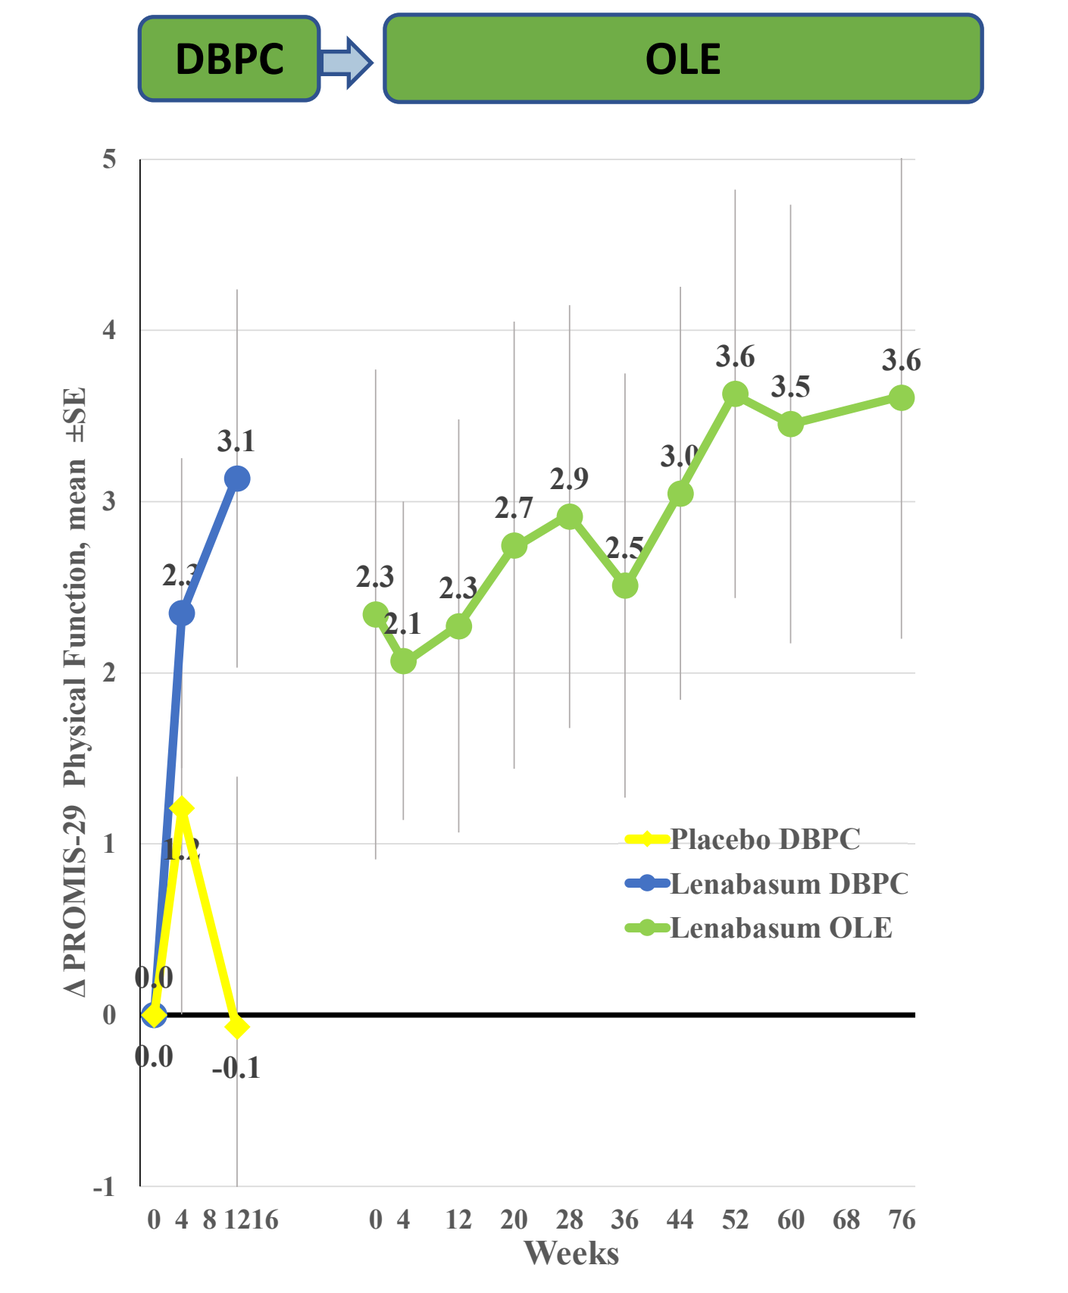

DBPC
OLE
Placebo DBPC
Lenabasum DBPC
Lenabasum OLE
Δ PROMIS-29 Physical Function, mean ±SE
Weeks

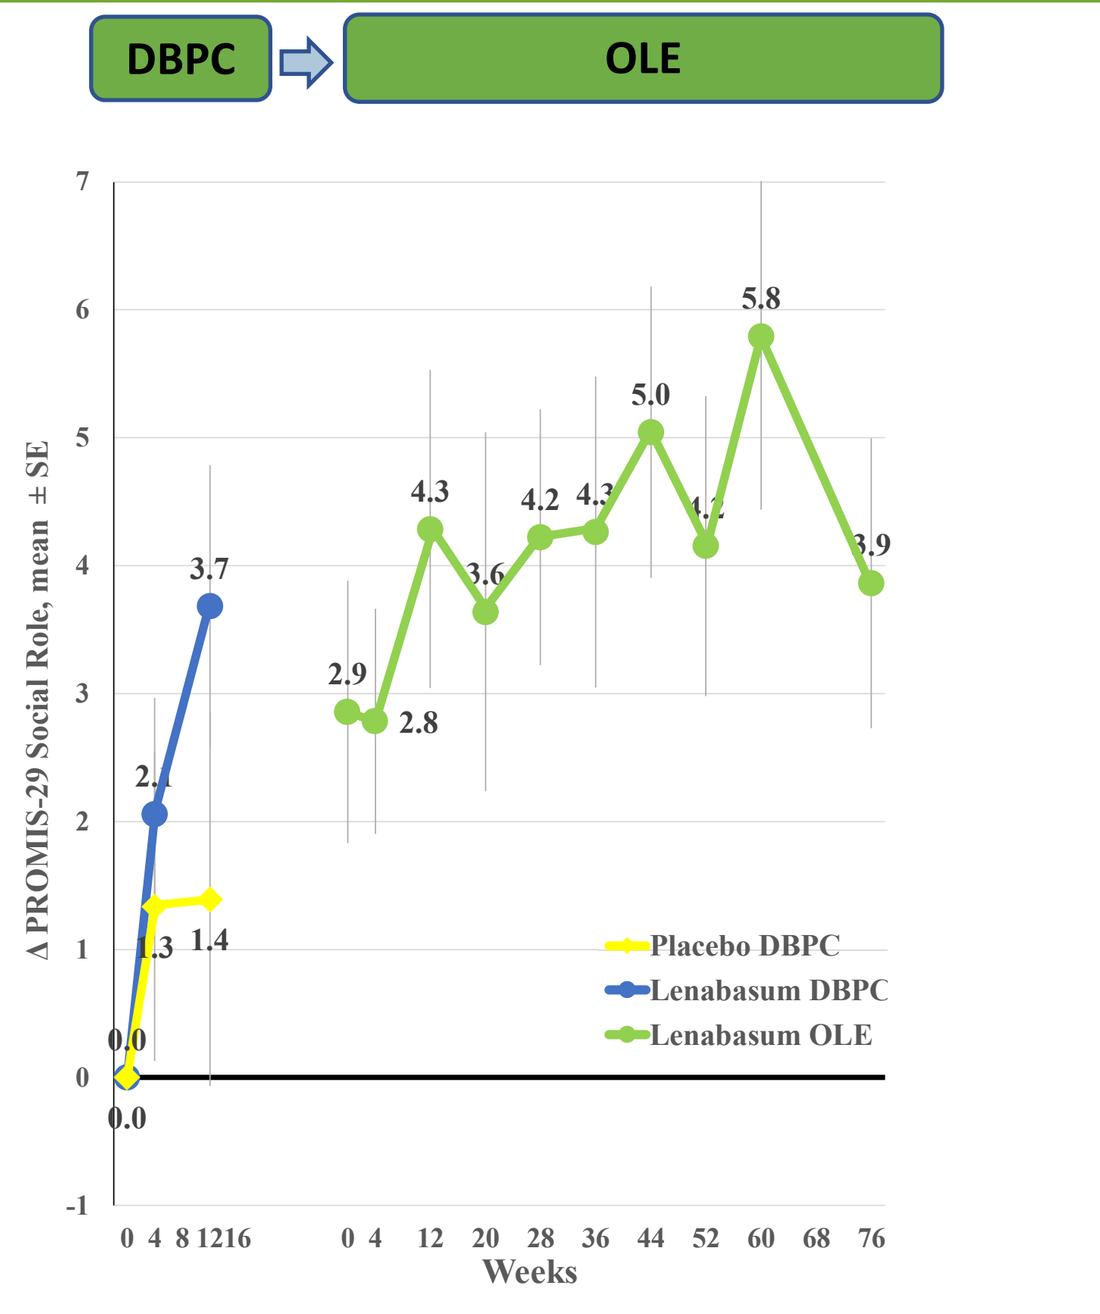

DBPC
OLE
Placebo DBPC
Lenabasum DBPC
Lenabasum OLE
Δ PROMIS-29 Social Role, mean ± SE
Weeks

- All three measures of patient-reported function improved during the OLE
- Mean improvement in HAQ-DI at 18 months exceeded reported 18-month MID of -0.10

Forced Vital Capacity

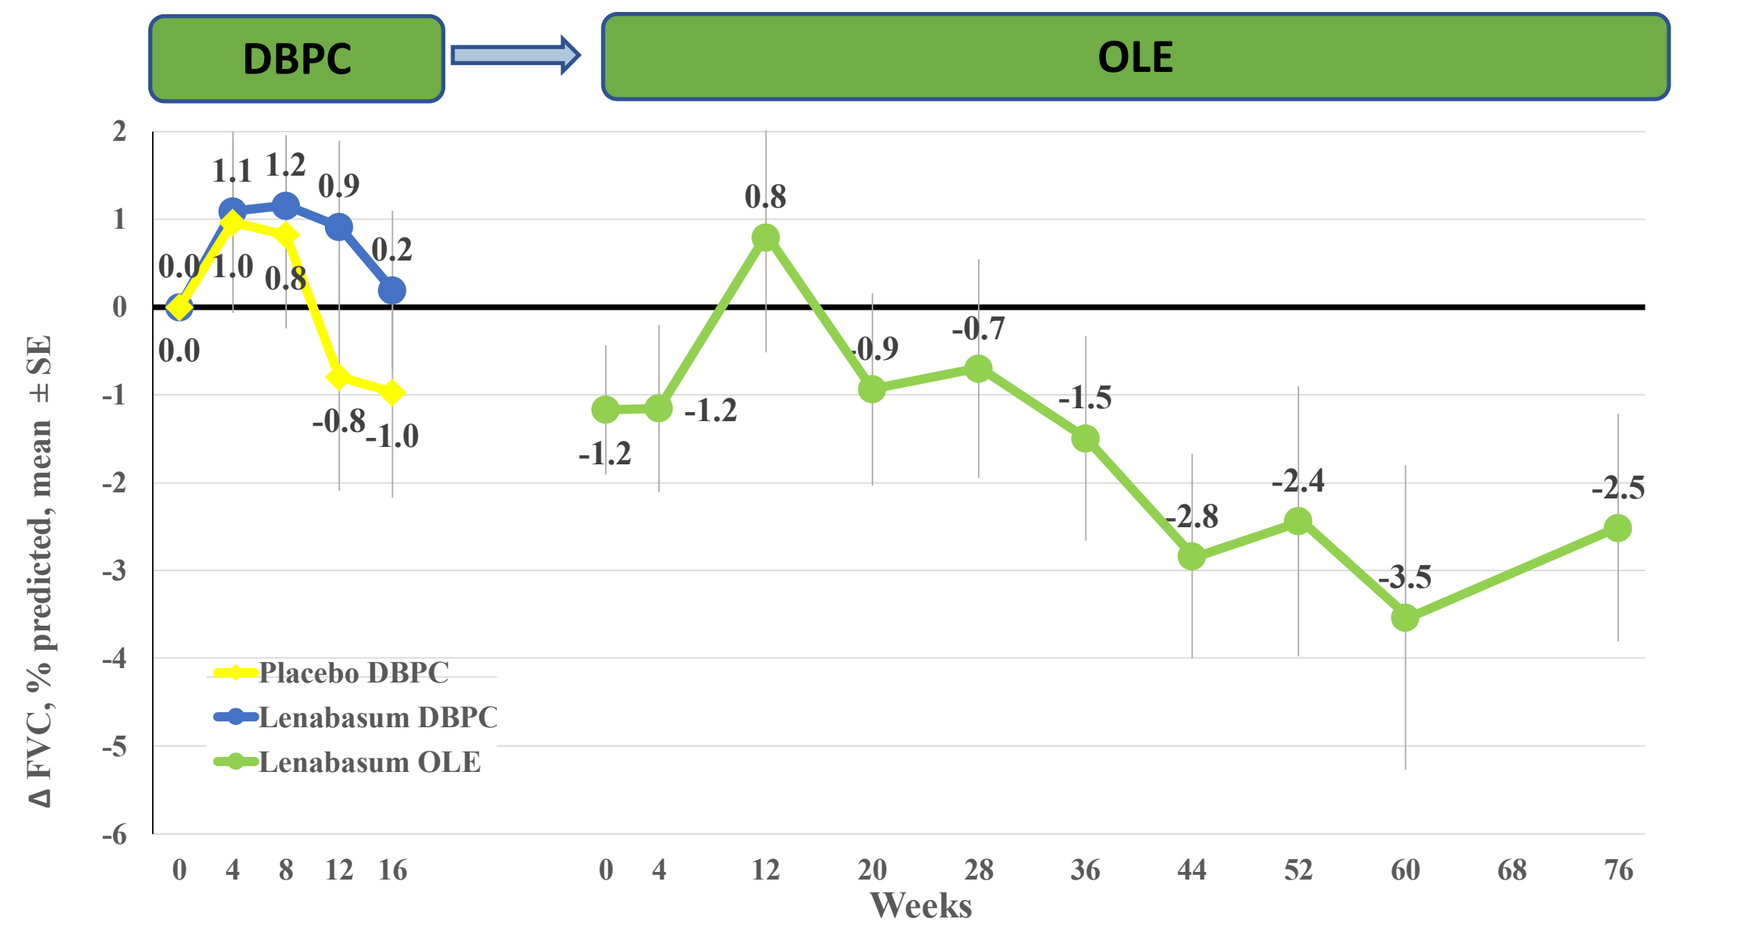

DBPC
OLE
Placebo DBPC
Lenabasum DBPC
Lenabasum OLE
Δ FVC, % predicted, mean ± SE
Weeks

- FVC % predicted stable from beginning of OLE through Month 9
- Small decrease in mean FVC % predicted seen after Month 9; compared to beginning of OLE, mean FVC % predicted decreased by 1.3% predicted at 12 months.

CONCLUSIONS

- Safety and tolerability profile of lenabasum in dcSSc remained very favorable after 18 months dosing in the OLE
- Subjects improved with lenabasum treatment in the OLE, as assessed by measures of overall disease, skin involvement, and patient function.
- Generally, improvement increased over time in the first 12 months of the OLE, then was stable thereafter, showing a durable response. A slight decrease was seen in FVC % predicted.
- The limitations of assessing efficacy with open-label dosing are acknowledged, as is the potential impact of any change in concomitant medications

THANK YOU

- **To the people with SSc who participated and are participating in this study**
- **To the investigators and study staff that are successfully executing this trial**
- **To our DSMB members Drs. Phillip Clements, Virginia Steen and Richard Silver**

This study was sponsored by Corbus Pharmaceuticals, Inc.