T Cell Immune Response to Multi-Indication Intralesional Immunotherapy for Malignant Melanoma

Vijay Ramiya[1], Michael Shamblott[1], Meghan Gentilini[1], Marina Bastawrous[1], Ashraf Dehlawi[1],
Maya Jerald[1], Kristina Hennessy[2], Tracy LaDue[3], Patricia Lawman[1] and Michael Lawman[1]

[1]Morphogenesis, Inc., Tampa, FL; [2]Hennessy Research Associates, Shawnee, KS; [3]Southeast Veterinary Oncology and Internal Medicine, Orange Park, FL.

INTRODUCTION

Melanoma is one of the most prevalent neoplasms seen in horses and occurs in 80% of gray horses over 15 years of age. Immunotherapy is a promising non-invasive and non-toxic treatment modality in veterinary and human melanomas, but requires the development of relevant immunoassays to gauge efficacy.

STUDY DESIGN & OBJECTIVES

The primary objective of this randomized double-blinded placebo-controlled study was to assess the safety and efficacy of the IFx-VET*Direct* immunotherapy in horses with confirmed malignant melanoma.

IFx-VET*Direct* is a double stranded, covalently closed, circular plasmid DNA (pDNA) designated pAc/*emm55*. pAc/*emm55* is a mammalian expression vector encoding the *emm55* gene derived from the bacterium *Streptococcus pyogenes*. Each dose of IFx-VET*Direct* contained 300 µg of pDNA suspended in Tris Ethylenediaminetetraacetic acid (TE). The regimen included four biweekly and two monthly injections. Direct injection of the pDNA into the tumor causes uptake by the tumor cells themselves and subsequent expression of Emm55 protein. The rationale for this approach is that expression of Emm55 protein by subject cells makes them appear foreign to the immune system and elicits a potent immune response directed against Emm55 and tumor neoantigens. The primary objectives of this study were to measure the safety and efficacy of IFx-VET*Direct* as a treatment for equine melanoma. Horses had no previous or concurrent treatment for melanoma.

A secondary objective of this study was to analyze the tumor antigen-specific T cell response. The goal was to develop methodology, document the induction of tumor specific immune responses and correlate the presence and magnitude of vaccine-induced immune responses to clinical outcomes. This study documented the *in vitro* stimulation of subject lymphocytes by tumor-specific antigens pre-vaccination **(week 1)** and eight weeks post final vaccination **(week 23)** as measured by the production of intracellular interferon gamma (IFNγ) and correlated the presence of vaccine-induced immune response to clinical outcomes (methodology outlined in Figure 1).

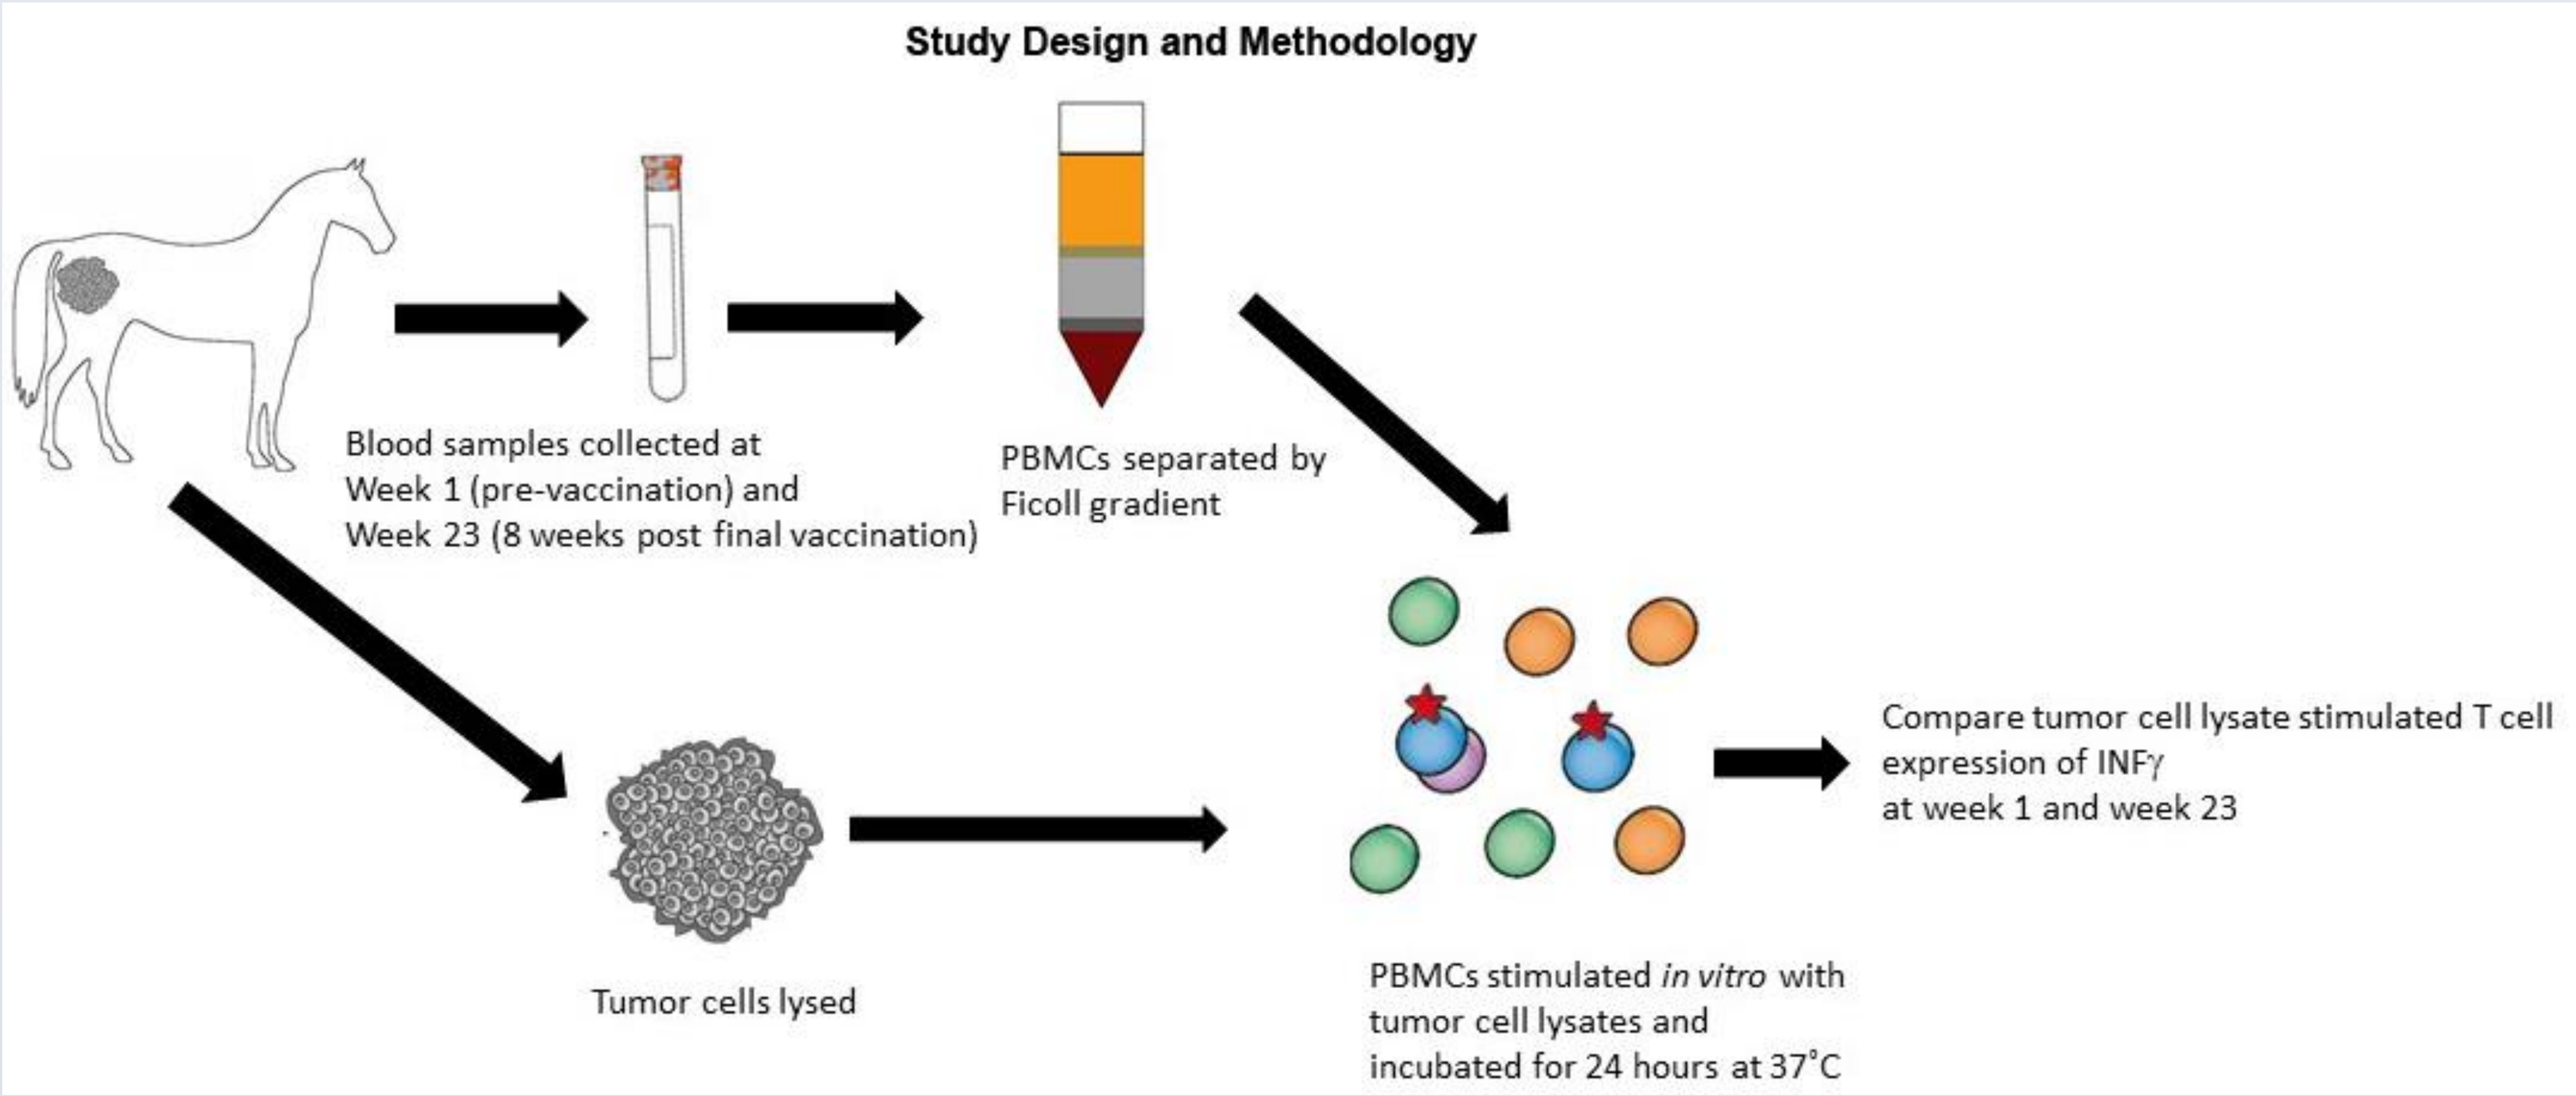
Study Design and Methodology
Blood samples collected at
Week 1 (pre-vaccination) and
Week 23 (8 weeks post final vaccination)
PBMCs separated by
Ficoll gradient
Compare tumor cell lysate stimulated T cell
expression of INFγ
at week 1 and week 23
Tumor cells lysed
PBMCs stimulated in vitro with
tumor cell lysates and
incubated for 24 hours at 37°C

Figure 1. Study design and methodology. Peripheral blood mononuclear cells (PBMC) collected pre-vaccination and eight weeks post last vaccination were stimulated with protein isolated from tumor cells.

RESULTS

It is widely accepted that IFNγ production by $CD4^+$ and $CD8^+$ T cells is critical for anti-tumor immunity. In this study, flow cytometry was used to measure IFNγ expression since it allows simultaneous identification of cell phenotype and cytokine expression. The result is presented as change in the absolute number of $CD4^+$ and $CD8^+$ T cells expressing IFNγ per mL of blood following tumor antigen stimulation at week 23 minus the change in expression at week 1. Statistical analysis for a linear association between tumor regression and IFNγ expression was carried out by linear regression. The disease control activity was defined as **the sum of animals with Partial Response (PR) and Stable Disease (SD), where PR is defined as ≥ 30% decrease in lesion size, Progressive Disease (PD) is defined as ≥ 20% increase and Stable Disease is defined as not PR or PD as per RECIST-VCOGv1.1 response criteria.**

- Of the 18 evaluable subjects, 16 received vaccine injections and two received control TE buffer (horse 10 and 21).
- Of the 16 vaccine recipients, the disease control activity (SD+PR) was 100%.
- Three subjects exhibited PR while 13 had SD.
- Both control subjects had SD. One exhibited 12.6% increase in tumor burden from baseline while the other exhibited an 8% regression.
- Of the 16 vaccinated subjects, 14 (87.5%) expressed increased $IFN\gamma^+$ cells per mL of blood, either of $CD4^+$ or $CD8^+$ T cells, upon antigen stimulation compared to pre vaccine levels.
- Of the two control subjects, horse 10 did not exhibit increase in the number of $IFN\gamma^+$ cells upon stimulation and horse 21 expressed a lower number of $IFN\gamma^+$ at week 23 than at week 1.
- There were no statistically significant relationships between percent change in tumor burden at week 23 and change in the number of $CD4^+$ $IFN\gamma^+$ (R^2=0.13; P=0.13) or $CD8^+$ $IFN\gamma^+$ (R^2=0.04; P=0.42) cells per mL of blood at week 23.
- There were no significant differences between week 1 and week 23 in the number of $CD4^+$ (P=0.09) or $CD8^+$ cells (P=0.67) cells per mL blood.

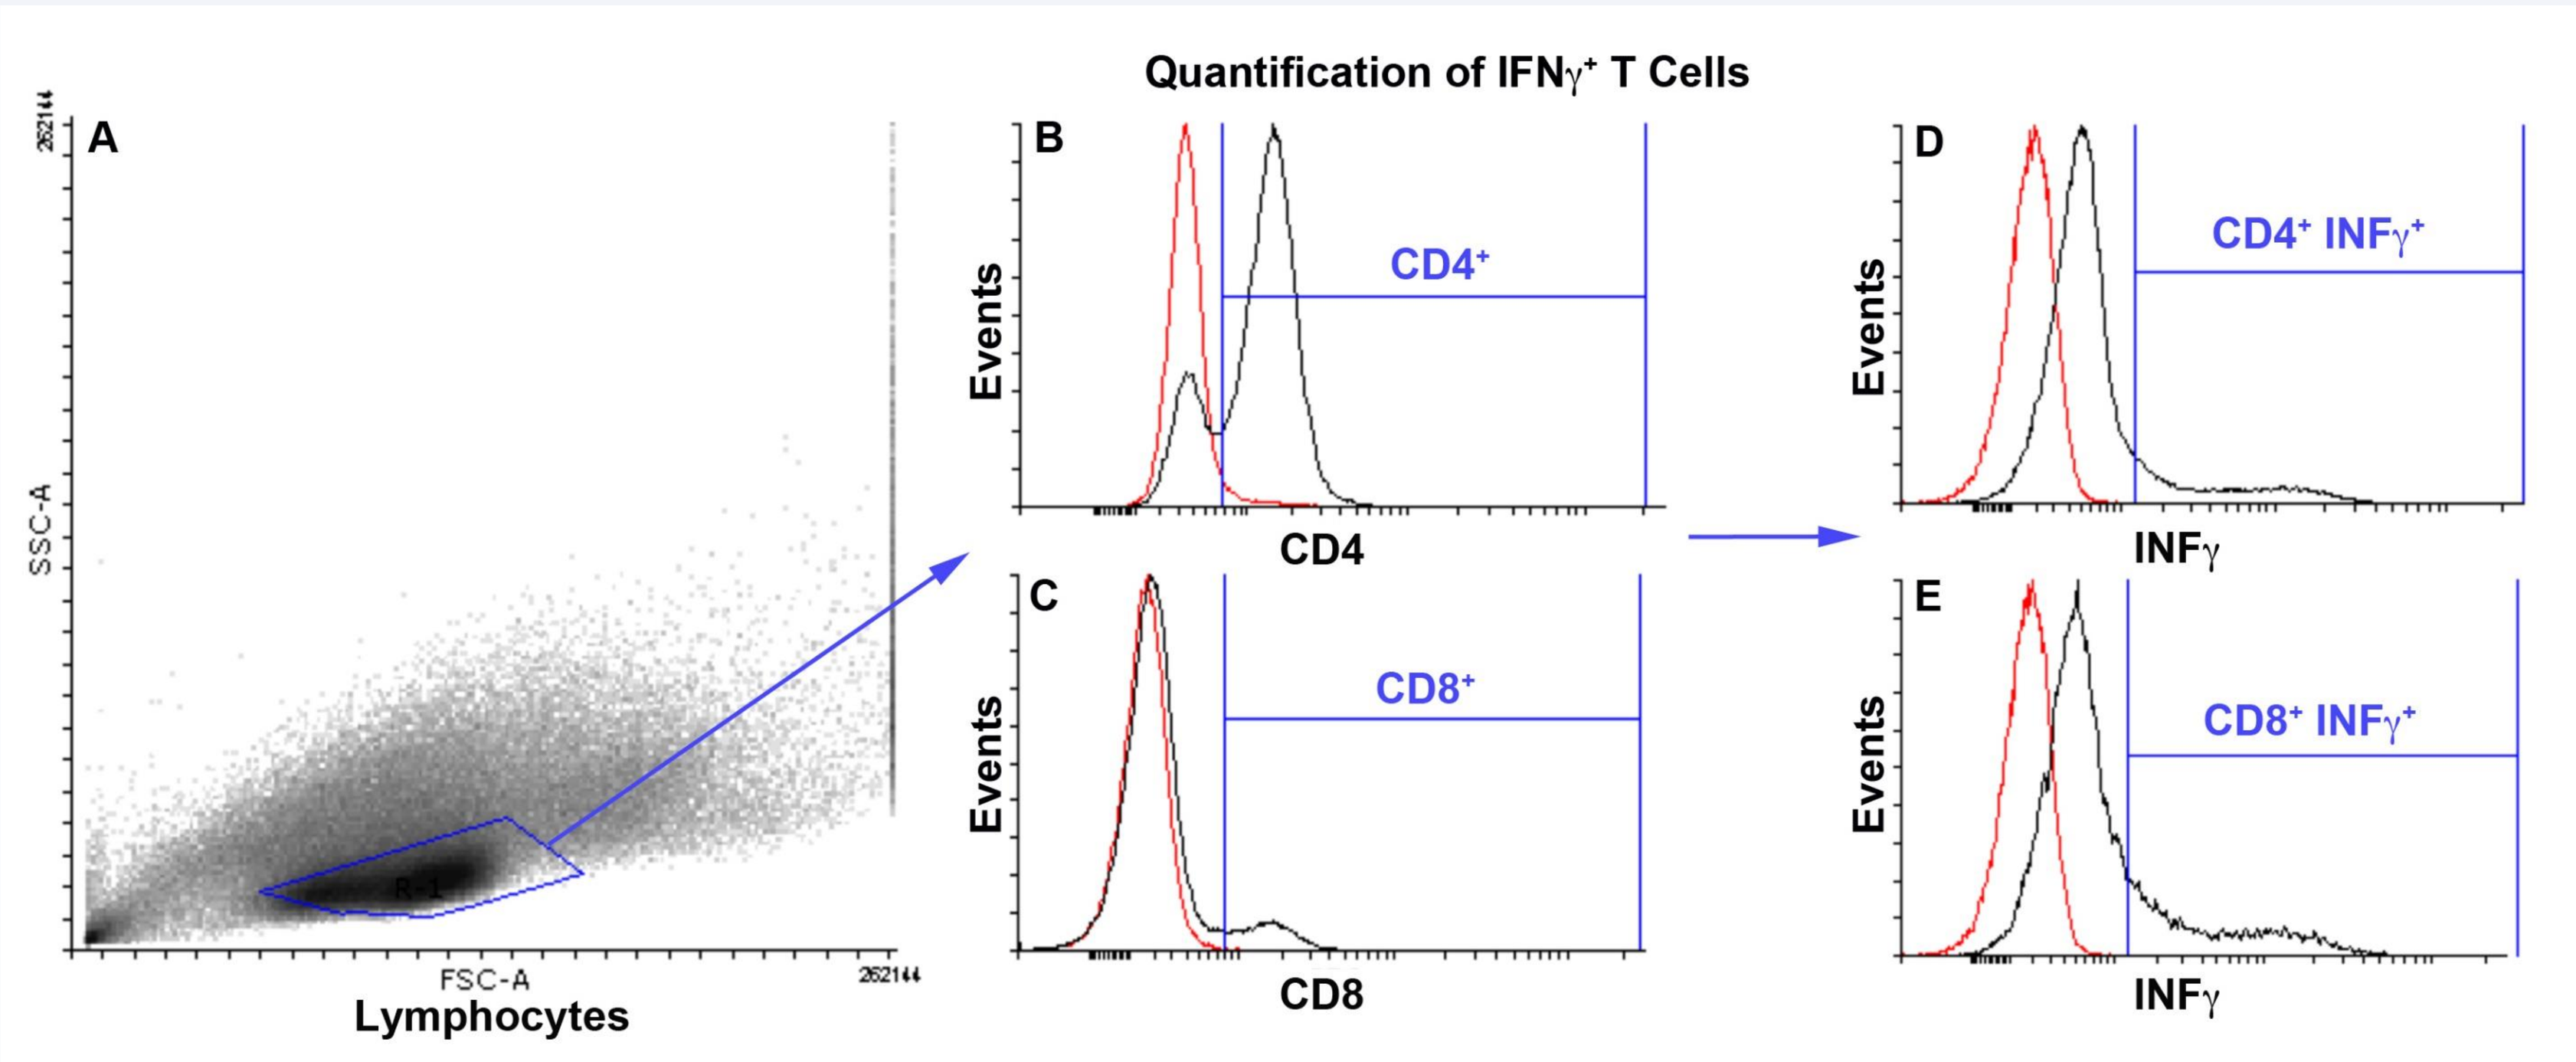
Quantification of IFNγ+ T Cells
A
SSC-A
FSC-A
Lymphocytes
B
Events
CD4+
CD4
C
CD8+
CD8
D
CD4+ INFγ+
INFγ
E
CD8+ INFγ+
INFγ

Figure 2. Quantification of T Cell expression of IFNγ by flow cytometry. A, Side scatter (SSC-A) and forward scatter (FSC-A) were used to identify lymphocytes in equine PBMC. **B-E**, red lines indicate cells stained with antibody isotype negative control, black lines indicate staining with antibody specified below each plot. Cells positive for each antibody fall within the blue gates. **B**,**C,** Helper T cells expressing CD4 and cytotoxic T cells expressing CD8 were identified by staining intensity within the blue gates. **D,E**, The percentages of $CD4^+$ and $CD8^+$ T cells that also express IFNγ were calculated based on the number of cells that fall within the positive gates.

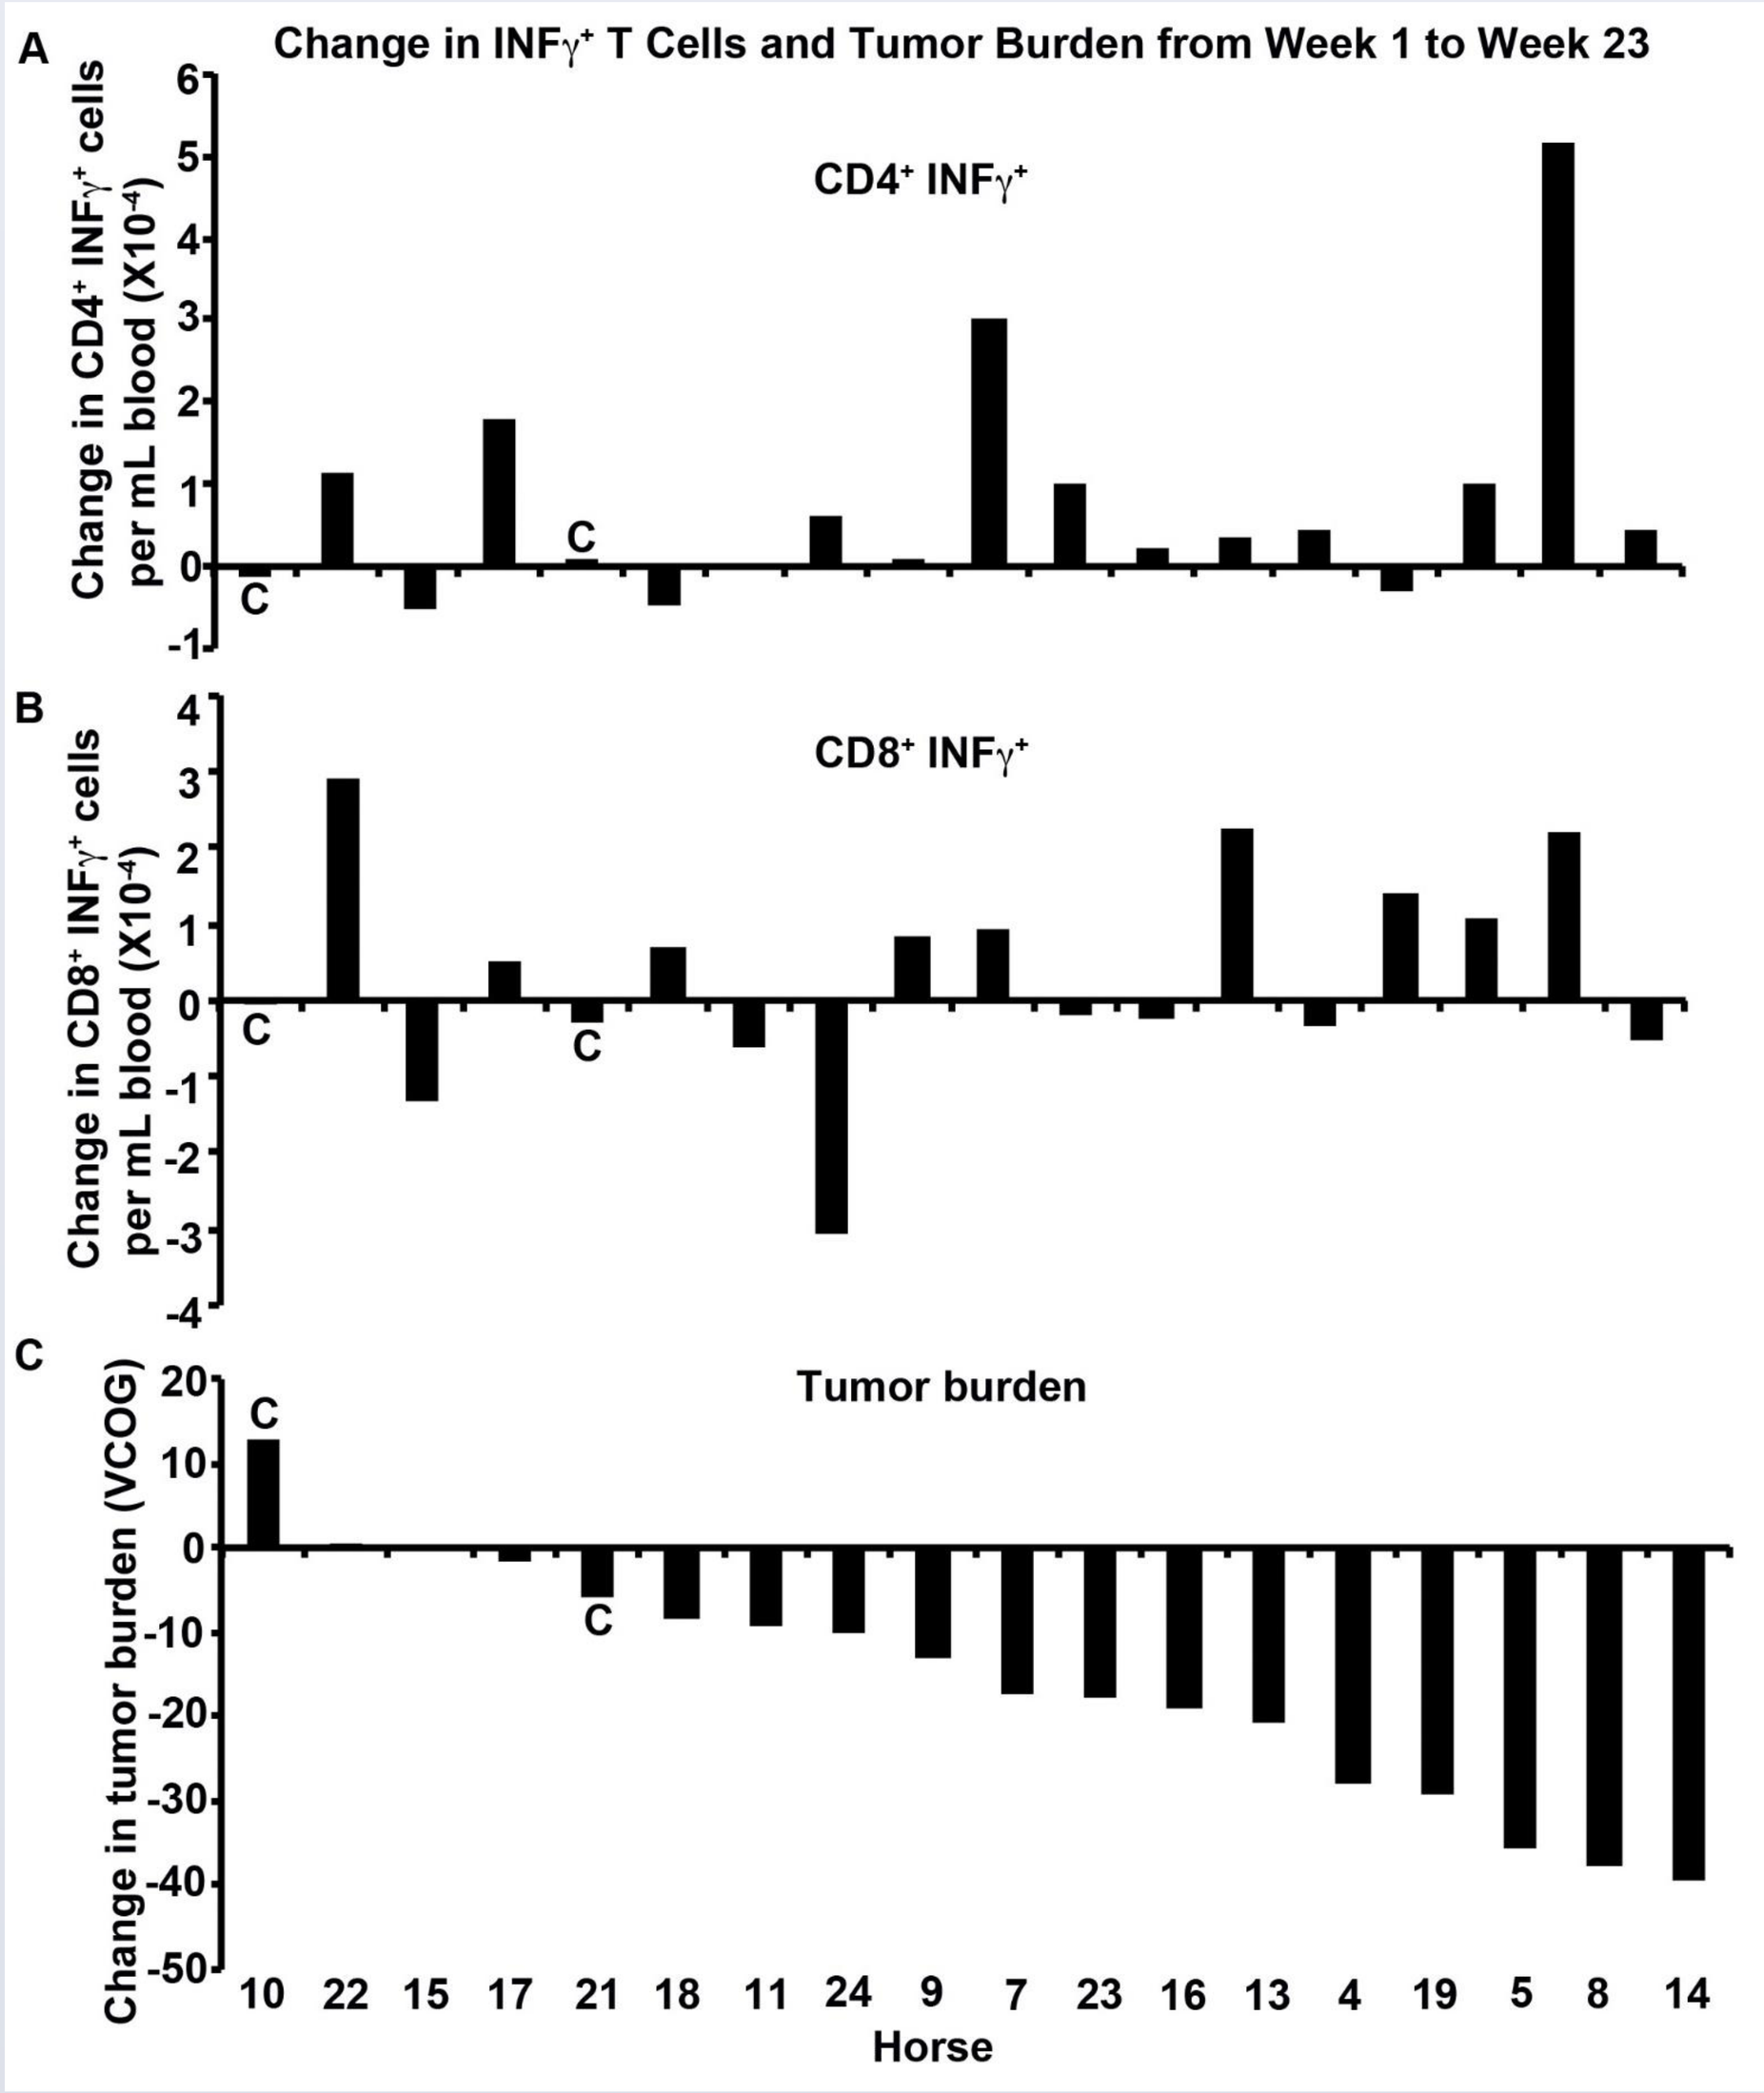
Change in INFγ+ T Cells and Tumor Burden from Week 1 to Week 23
A
CD4+ INFγ+
Change in CD4+ INFγ+ cells per mL blood (X10-4)
B
CD8+ INFγ+
Change in CD8+ INFγ+ cells per mL blood (X10-4)
C
Tumor burden
Change in tumor burden (VCOG)
10 22 15 17 21 18 11 24 9 7 23 16 13 4 19 5 8 14
Horse

Figure 3. Change in $IFN\gamma^+$ T cells and tumor burden from week 1 to week 23. Tumor antigen-stimulated IFNγ production by equine T cells indicated as change in absolute numbers of **A**, $CD4^+$ $IFN\gamma^+$ and, **B,** $CD8^+$ $IFN\gamma^+$ cells per mL of blood. A negative value means no additional increase upon stimulation with antigen between week 1 and 23. **C**, Percent change in tumor burden from baseline was determined at week 23 following RECIST-VCOGv1.1 guidelines. Control horses receiving TE injection are indicated by C above or below the plotted value.

CONCLUSIONS

- A valid assay to measure antigen-specific cytokine production has been established for horses.
- Wide immune response variability was expected due to small sample size and heterogeneity of outbred populations.
- IFNγ results demonstrate a durable tumor-specific vaccine-induced immune response in vaccinated horses 168 days post vaccine initiation.
- Week 23 (8 weeks post-final vaccination) may represent the stable or memory phase of the cellular response.

The positive outcome of this equine melanoma study and universality of priming anti-tumor immune response make this direct **multi-indication** intralesional immunotherapy a cost effective approach to veterinary cancers; **equine, canine and feline.**

ABOUT MORPHOGENESIS

Morphogenesis is a clinical stage company developing novel cell and gene therapies based on distinct and synergistic technology platforms. Our mission is to change the way chronic diseases are treated by engaging the innate intelligence of the body.

Veterinary Oncology Services, Inc. is a fully integrated service provider of personalized IFx-VET cancer vaccines for companion animals. In 2013, the Company gained authorization from the USDA to provide the personalized service across the US. Customization of the technology for each specific patient has been made possible by carefully honing the process so that it is rapid, efficient and cost effective. The Veterinary Oncology Services (VOS) process has been tested in laboratory mice and in companion animals in clinical settings. To date, personalized IFx-VET vaccines have been provided to veterinarians for the treatment of 37 different types of cancer in over 113 breeds of dogs, cats and horses with extremely favorable results. IFx-VET is an entirely new and effective weapon for veterinarians to use in their fight against cancer in companion animals. VOS is headquartered in Tampa, FL and provides services to veterinarians and their clients throughout the US. For more information please visit www.ifx-vet.com.

RESEARCH POSTER PRESENTATION DESIGN © 2015
www.PosterPresentations.com

Up until the printing of this poster the USDA has not reviewed these data or licensed this biologic.