Phase 1 Trial: Intralesional Immunotherapy with IFx-Hu2.0 Vaccine in Patients with Advanced Merkel Cell or Cutaneous Squamous Cell Carcinoma

Ashraf Dehlawi[1], Christopher Konig[1], Michael Lawman[1], Patricia Lawman[1], Lucy DesJardin[1], Andrew Brohl[2], Kenneth Tsai[2]

1. Morphogenesis, Inc., 10500 University Center Drive, Suite 110, Tampa, FL 33612; 2. Moffitt Cancer Center, 12902 USF Magnolia Drive Tampa, FL 33612

ABSTRACT

Growing evidence over the past decade suggests that active immunotherapy is a viable and clinically effective treatment option for patients with Merkel cell carcinoma (MCC) or cutaneous squamous cell carcinoma (cSCC) experiencing some response to immunotherapy. The objective response (tumor shrinkage) rate to systemic biological response modifiers such as anti-PD-1 antibodies is approximately 56%. However, an unmet medical need for improving patient treatment response rates remains.

The novel therapeutic plasmid DNA vaccine, IFx-Hu2.0, stimulates a patient-specific immune response targeting cancer cells. IFx-Hu2.0 exploits the inherent vulnerability of the antigenic signature present to create a broad immune response regardless of underlying complexity and variability. It introduces the *emm55* gene from *Streptococcus pyogenes* to cancer cells, which in turn express the Emm55 protein. This prominent signal initiates an immune cascade leading to the activation of multiple immune cells, including cytotoxic T cells.

While pre-clinical murine melanoma data showed enhanced anti-tumor activity with IFx-Hu2.0 + anti-PD-1 combination therapy, pre-clinical equine melanoma data (n=22) showed a disease control rate of 100% with pAc/*emm55* (drug substance) monotherapy alone (disease regression=77%; stable disease=73%; partial response=27%).

BACKGROUND

Proof-of-concept studies were designed to assess the safety and efficacy of IFx-Hu2.0 in mice. In summary, a total of 10 experiments were conducted to assess various aspects of the vaccine's mechanism of action, efficacy, safety and biodistribution. A total of 257 mice were evaluated in these experiments with treatment group outcomes showing significantly favorable results as compared to control groups. In addition, the assessments of safety and toxicity supported proceeding to first-in-human studies.

In addition, the drug substance (plasmid DNA pAc/*emm55* in TE buffer) was evaluated for safety and efficacy under a USDA sanctioned field safety and efficacy study as an aid in the treatment of melanoma in equines.

IFx-Hu2.0 will be studied in two separate non-randomized, open-label, and uncontrolled phase 1 trials. A currently active trial (CM 2017-01/MCC #19500) is evaluating IFx-Hu2.0 as a monotherapy in patients with unresectable stage III/IV cutaneous melanoma (CM), while a planned trial will evaluate the same drug product as a monotherapy in patients with advanced Merkel cell carcinoma (MCC) or cutaneous squamous cell carcinoma (cSCC) (NMSC 2019-01/MCC #20162).

PRECLINICAL STUDIES

IFx-Hu2.0 increases T cell infiltration of tumors

Figure 1: IFx-Hu2.0 intralesional injection into B16 melanoma tumors increases CD8+ T cell infiltration. B16 melanoma tumors were harvested and the numbers of CD8+ T cells increased with injection of the emm55 vector as measured via the mean fluorescence intensity (MFI) in flow cytometry ($p < 0.05$, unpaired T test).

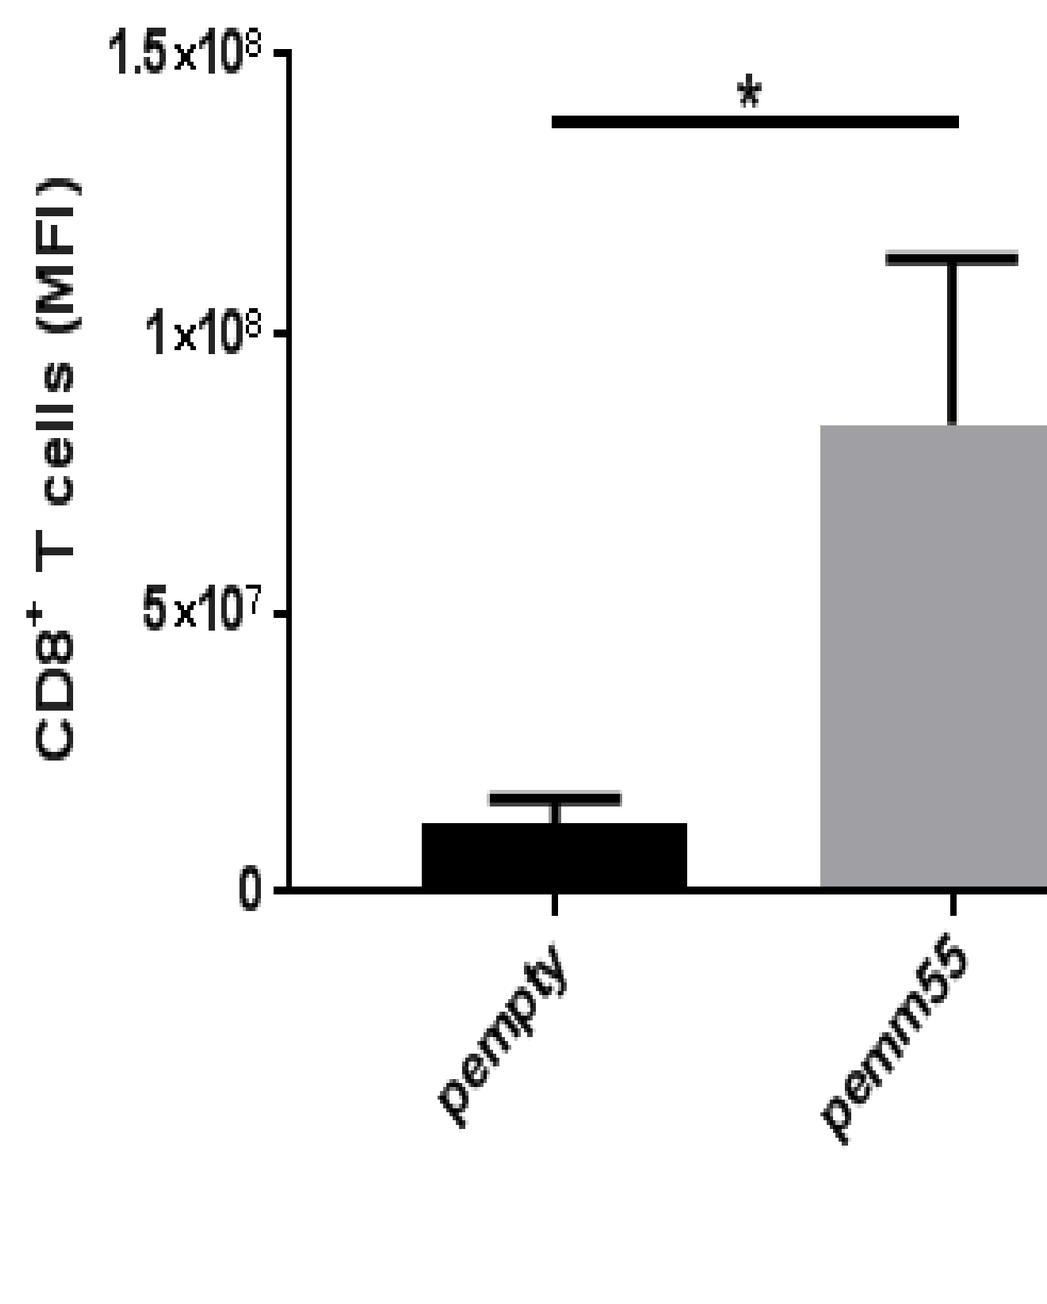

CD8+ T cells (MFI)
1.5x10^8
1x10^8
5x10^7
0
*
pempty
pemm55

RESEARCH POSTER PRESENTATION DESIGN © 2019
www.PosterPresentations.com

IFx-Hu2.0 decreases tumor mass in a murine model

Figure 2: IFx-Hu2.0 reduced the weight of B16 melanoma tumors. Tumor weight significantly decreased after injection of the emm55 vector into B16 murine tumors (N = 6 per group, p = 0.0076 unpaired T test).

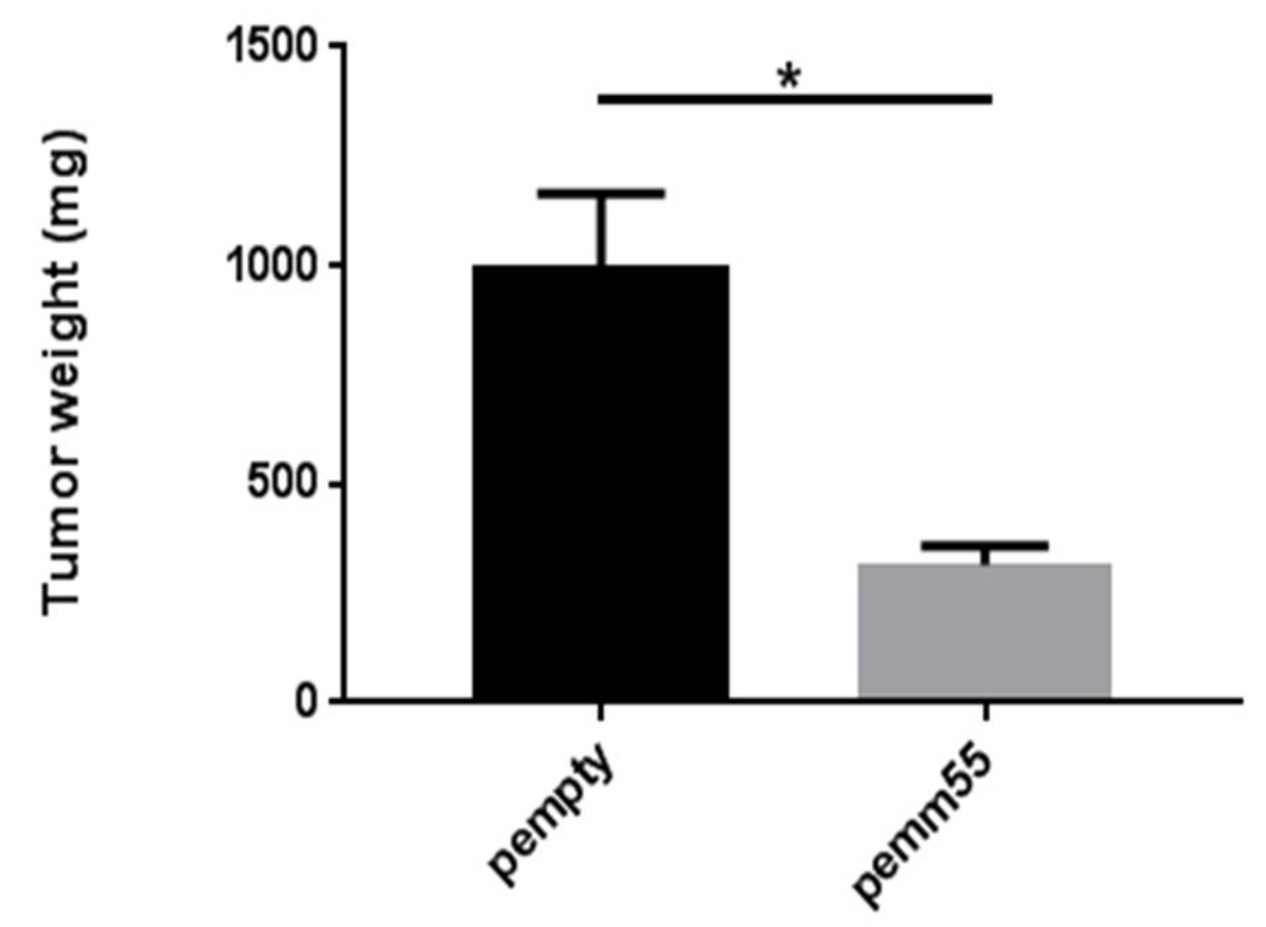

Tumor weight (mg)
1500
1000
500
0
*
pempty
pemm55

IFx-Hu2.0 increases T cell infiltration of tumors

Figure 3 (right): Immune cell infiltration of the tumor after treatment with emm55. Intralesional injection of pAc/emm55 into B16 melanoma tumors increases T cell infiltration. We observed trends in the numbers of infiltrating CD3+ and CD8+ cells with CD4+ cells reaching statistical significance p = 0.049.

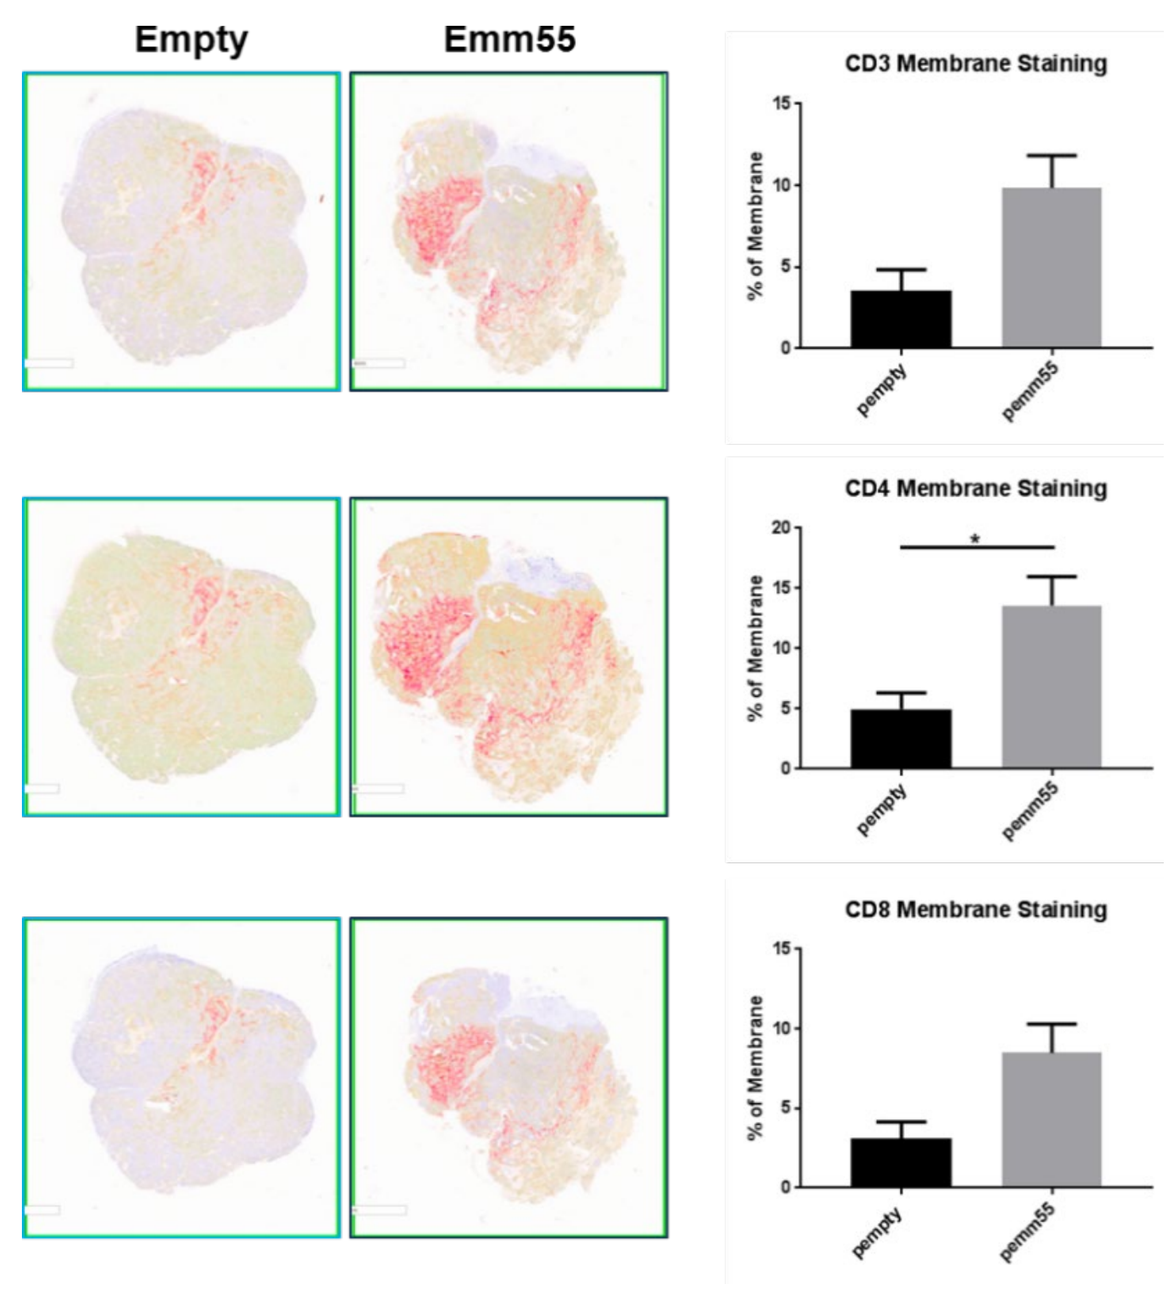

Empty
Emm55
CD3 Membrane Staining
CD4 Membrane Staining
CD8 Membrane Staining
% of Membrane
pempty
pemm55

IFx-Hu2.0 activity is CD4+ and CD8+ T cell dependent

Figure 4: Systemic immune responses elicited by the emm55 vector in B16 bearing mice is T cell dependent . C57BL/6 mice were injected with anti-CD4 or anti-CD8 antibody (300 μg/200μL/intraperitoneal) starting on day -3 and twice a week until the endpoint. Control mice received isotype control. On day 0, the mice received the B16 cells (1*105/100 μL/mouse/subcutaneously), N = 6 per group. IFx-Hu2.0 was injected weekly. An unpaired T test was utilized to compare the sizes of the tumors at the end of the experiment. There are clearly at least strong trends to support CD4+ and CD8+ mediated effects.

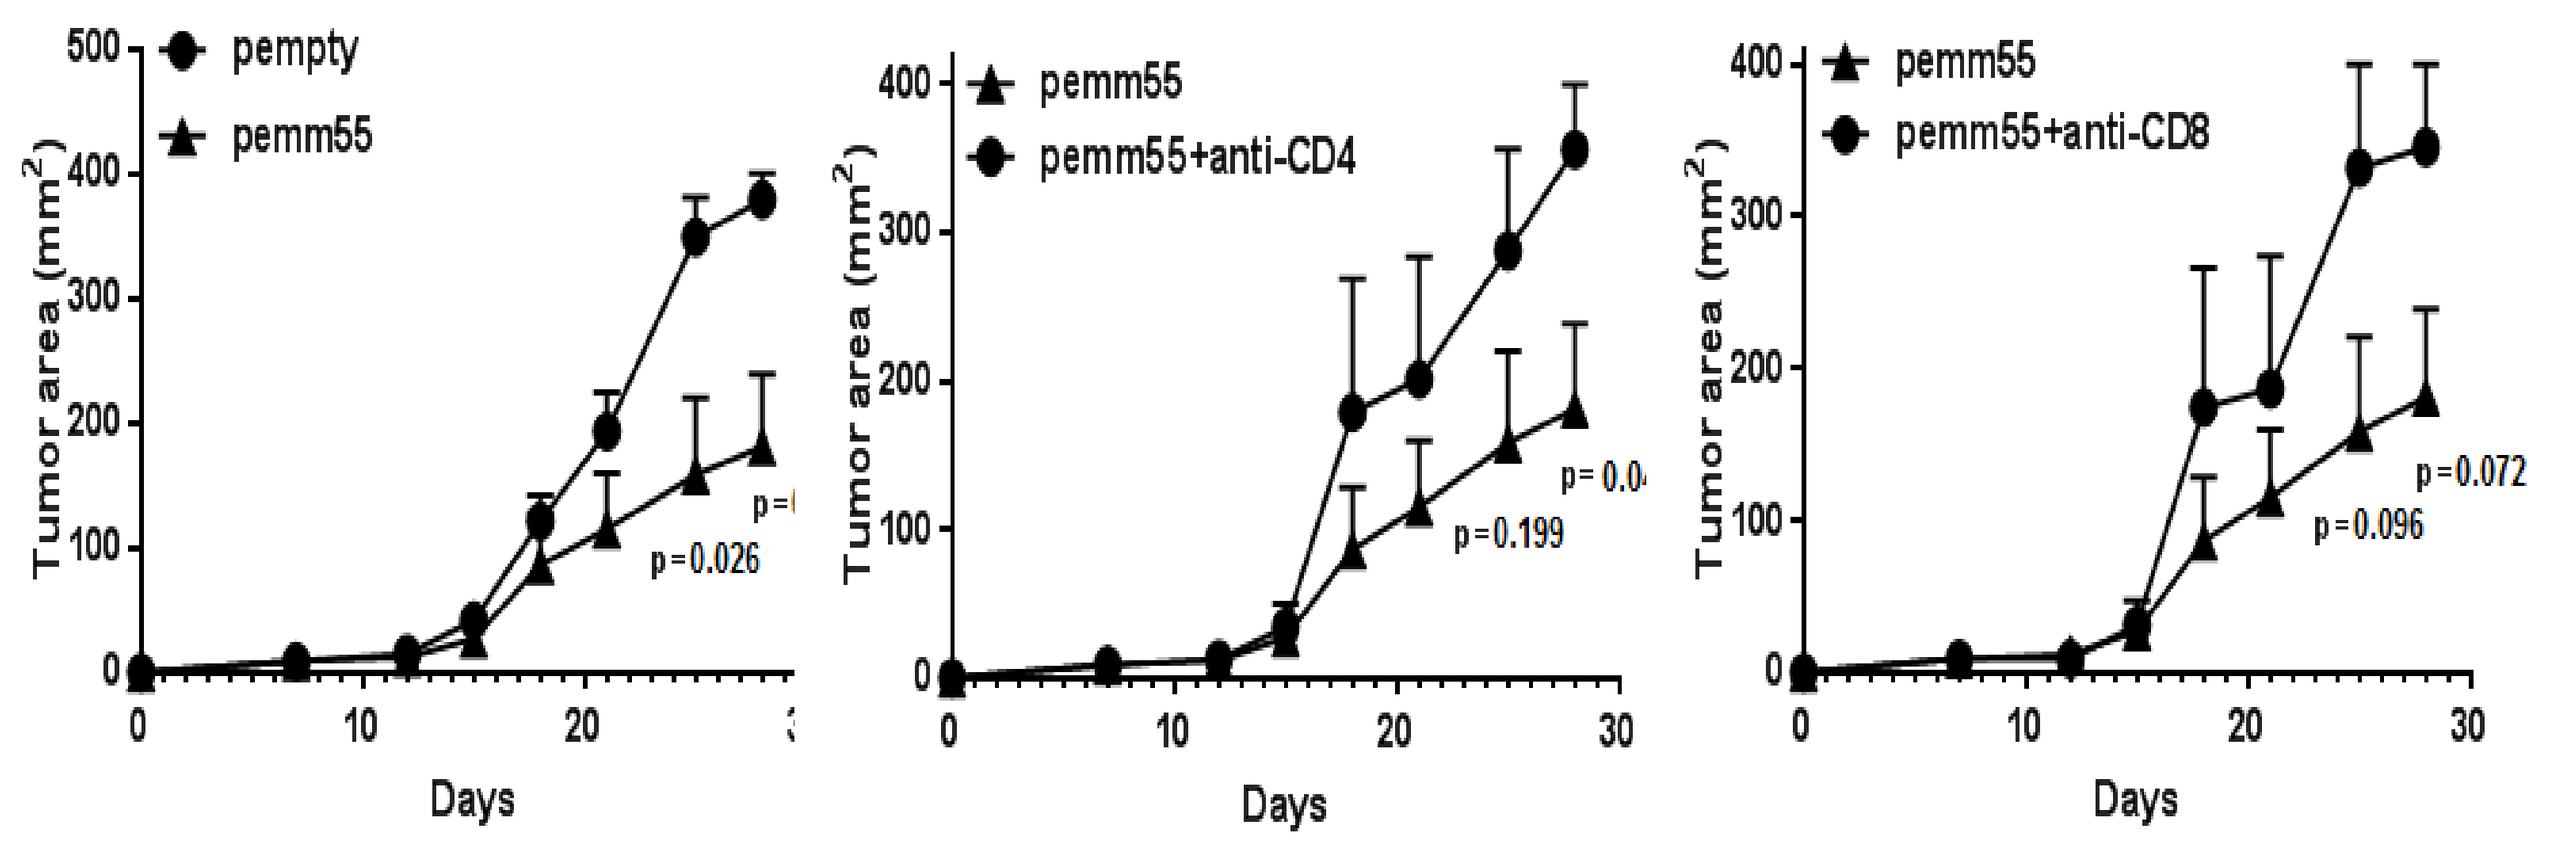

pempty
pemm55
pemm55+anti-CD4
pemm55+anti-CD8
Tumor area (mm2)
Days
p=0.026
p=0.199
p=0.096
p=0.072

IFx-Hu2.0 activity is antigen specific

Figure 5: Systemic immune responses elicited by emm55 vector in B16 bearing mice are CD4^{+} and CD8^{+} T cell dependent and are antigen specific. C57BL/6 were injected subcutaneously with M05 cells and the mice received intralesional injection of empty or emm55 vector on days 7 and 14. In addition, the mice received 5*10^{6} OT-1 T cells. After 48 hours, tumors were harvested and tetramer specific CD8^{+} T cells were measured by flow cytometry. The bar graphs represents the percent of positive cells (n=5 in each group).

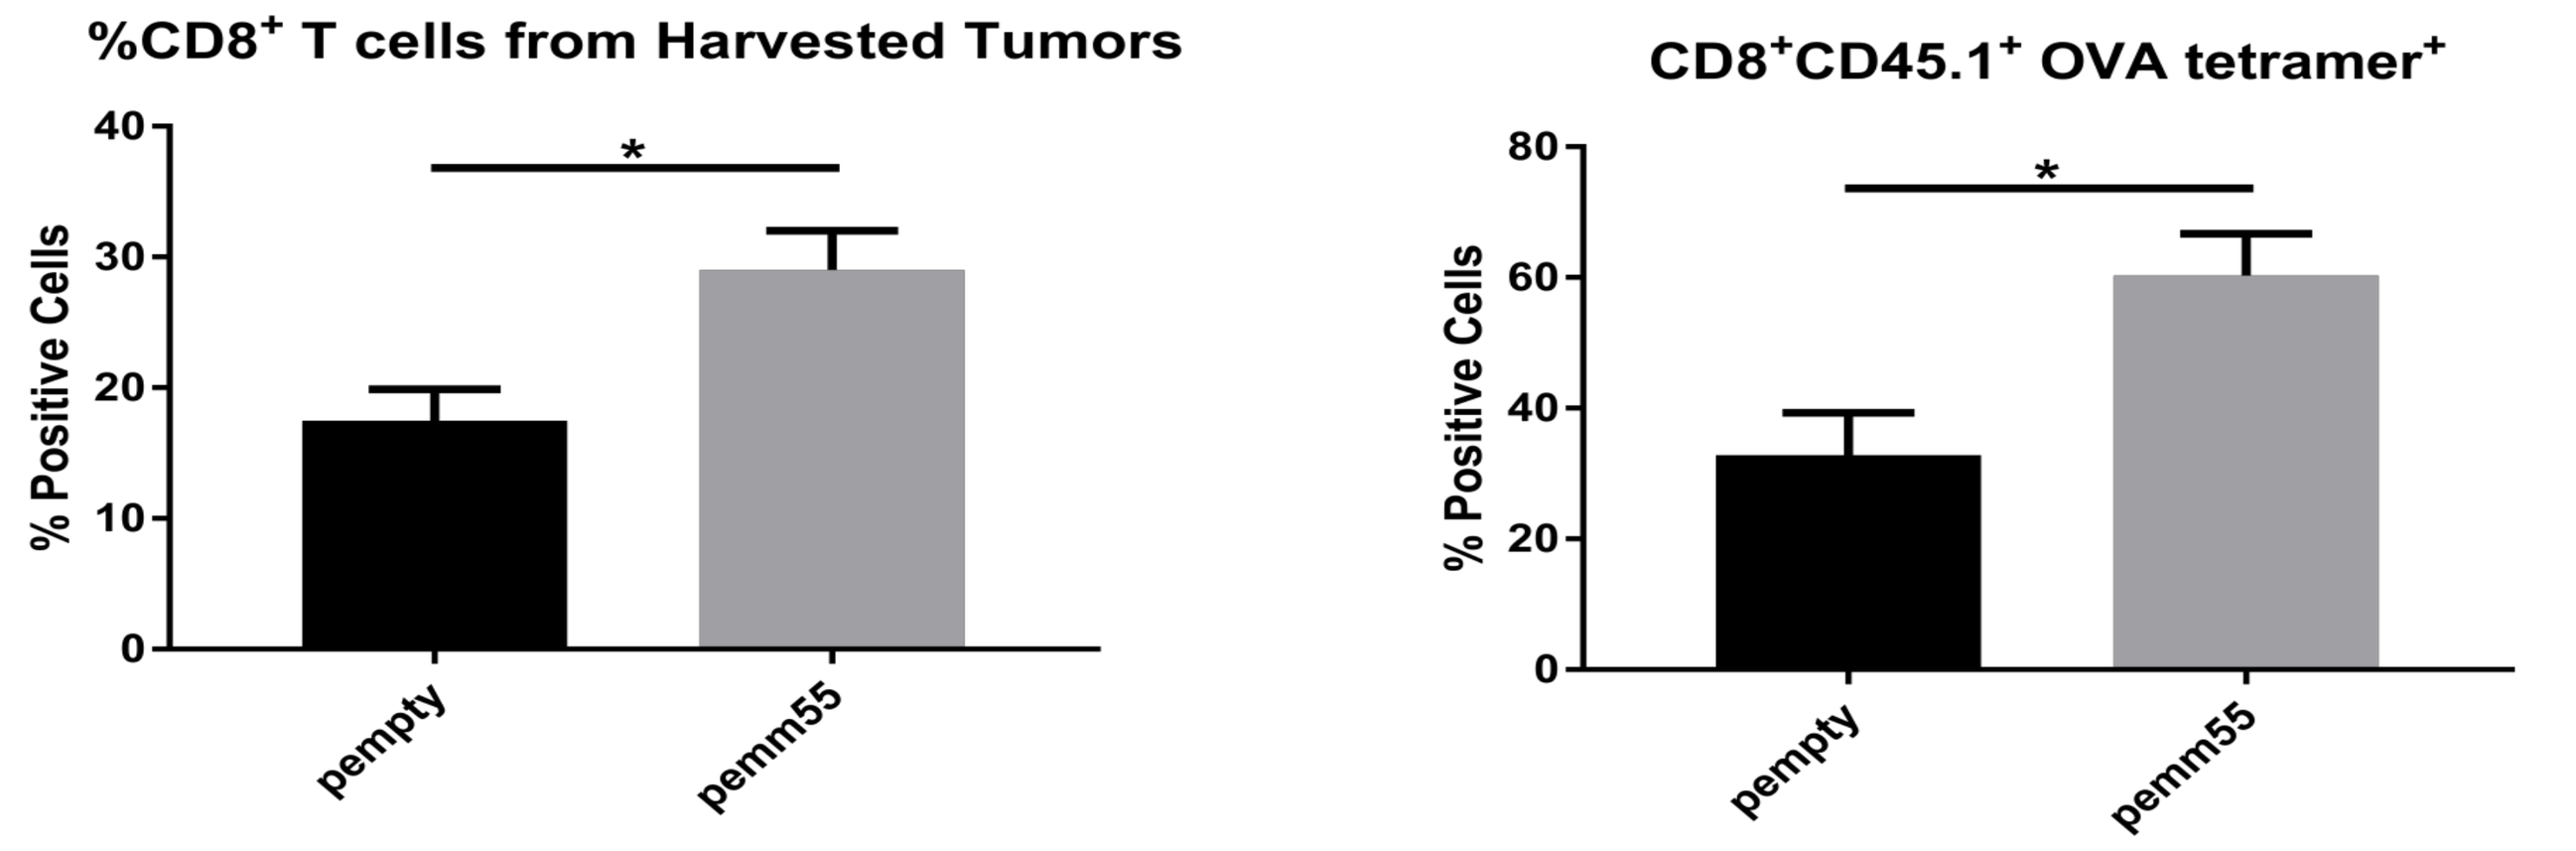

%CD8+ T cells from Harvested Tumors
CD8+CD45.1+ OVA tetramer+
% Positive Cells
*
pempty
pemm55

IFx-Hu2.0 ± anti-PD-1 therapy increases IFNγ production

Figure 6: IFNγ production increases upon the addition of emm55 vector and anti-PD-1 therapy. Murine melanoma B16 cells were injected subcutaneously on day 0. Empty vector or emm55 vector was injected on days 7,14, and 21. Starting on day 8, mice received intraperitoneal injection of either isotype control antibody or anti-PD-1 blocking antibody (250 μg/200μL/IP twice a week in combination with intralesional injections of empty or emm55 vector (N=12 for treatment groups, N=9 for control pEmpty group) Left Panel: Splenocytes collected from mice treated with emm55 vector ± anti-PD-1 therapy were co-cultured with B16 melanoma cells and culture supernatants were collected after 24 hours. Levels of IFNγ were measured ($p < 0.01$ utilizing unpaired T test with Bonferroni correction). Right Panel: Tumor sizes for these B16 tumors at day 25 were also measured and demonstrated an increased response with IFx-Hu2.0/anti-PD-1 compared to anti-PD-1 therapy alone ($p < 0.01$; unpaired T test).

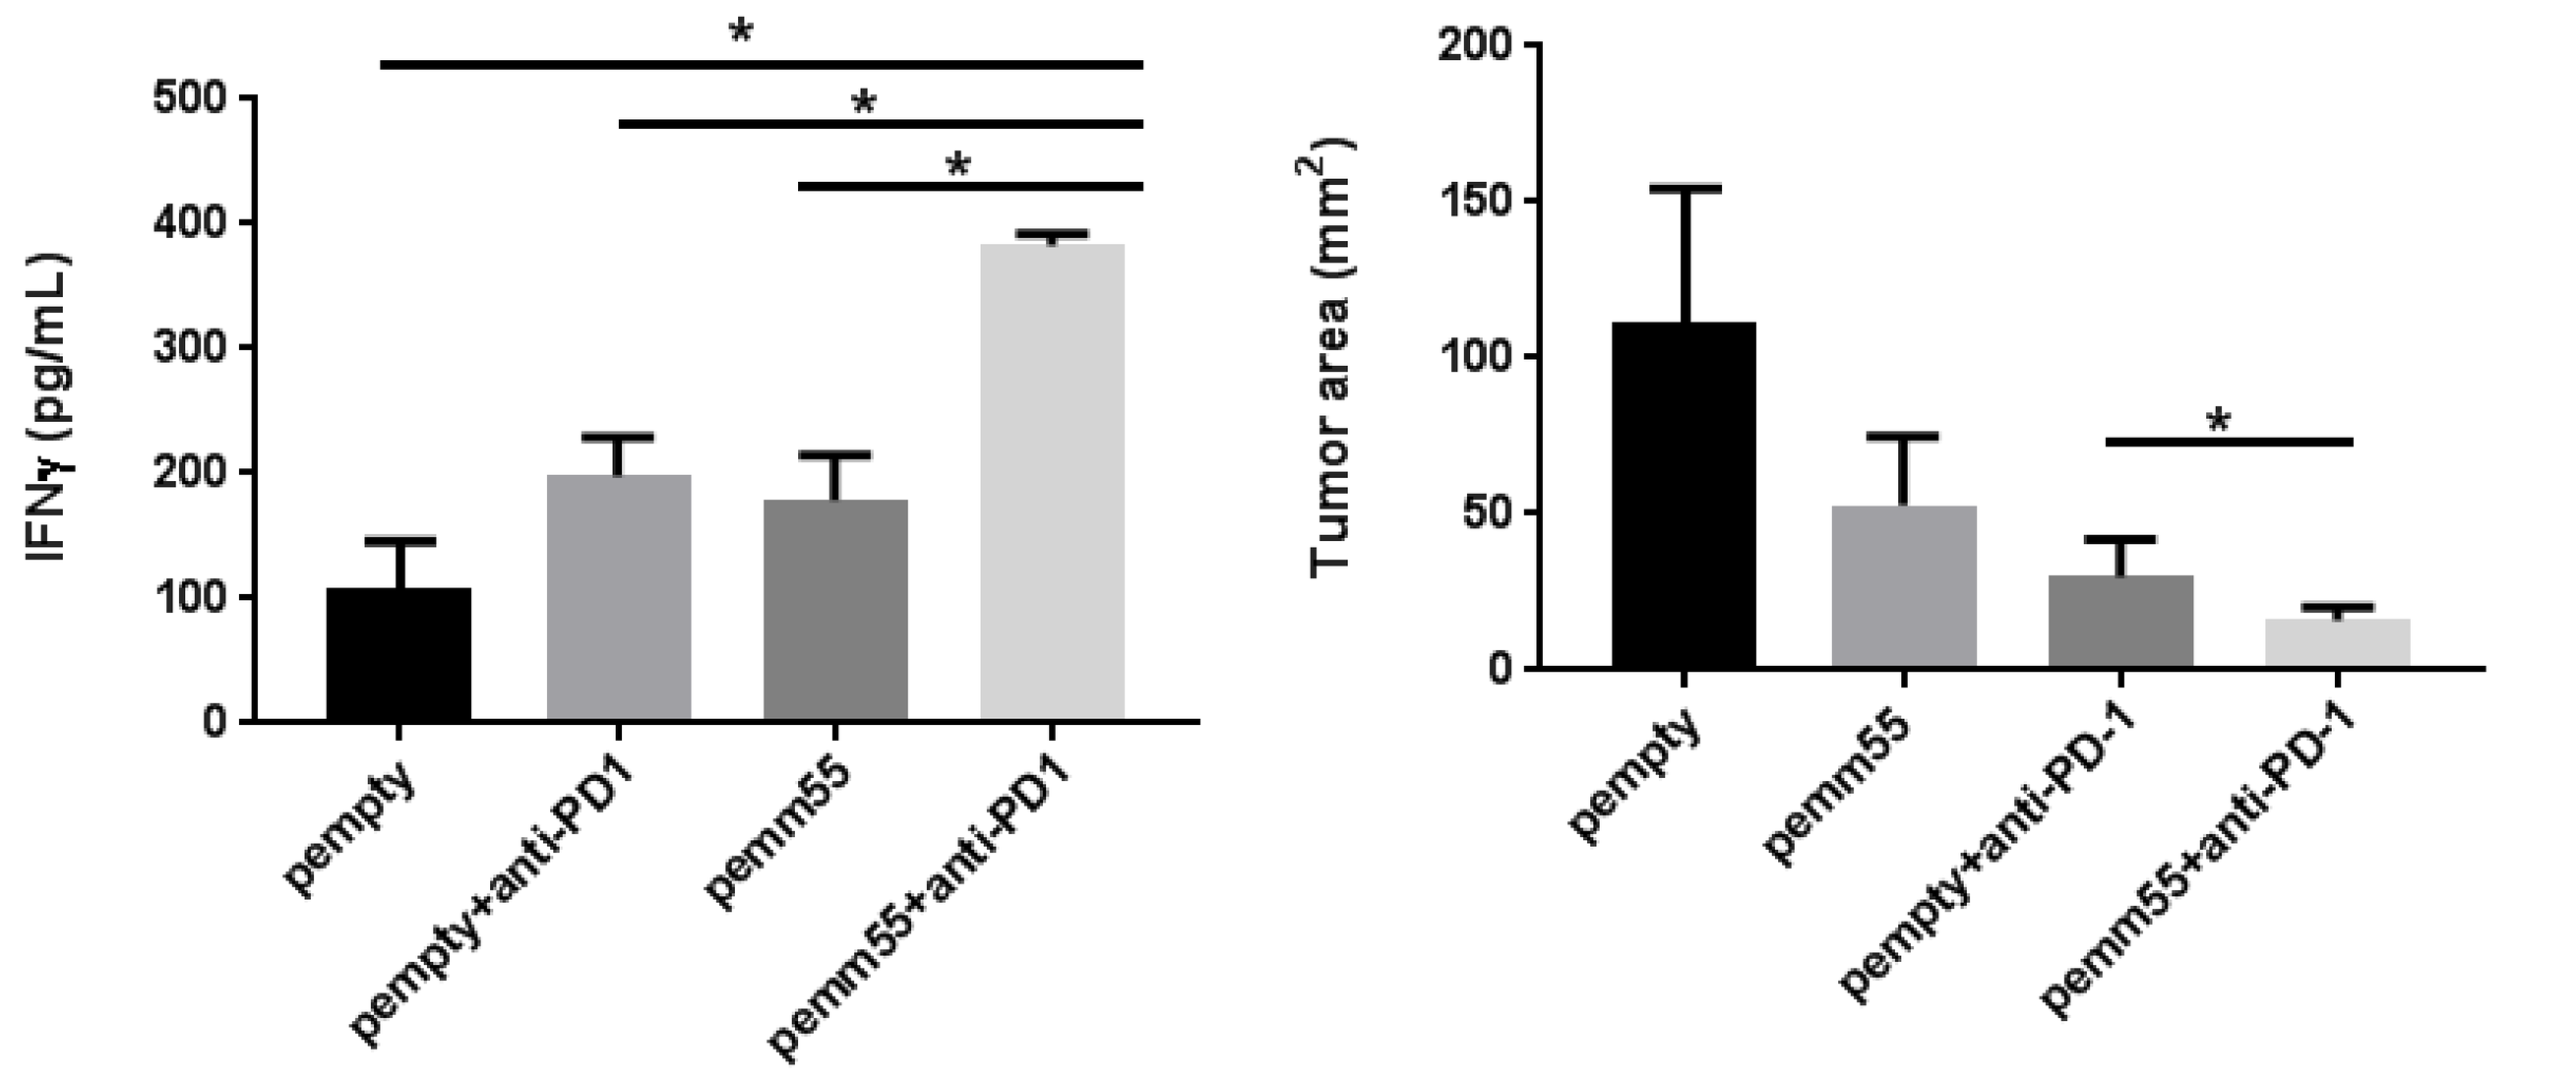

IFNγ (pg/mL)
Tumor area (mm2)
pempty
pempty+anti-PD1
pemm55
pemm55+anti-PD1
pempty+anti-PD-1
pemm55+anti-PD-1

Study of drug substance (plasmid DNA pAc/*emm55* in TE buffer) in naturally occurring melanoma in equines

Figure 7: The drug substance was injected into equine melanoma subjects and the equine subjects experienced responses to the intralesional therapy. This study was utilized to support the IND application for the first in human trial utilizing the drug substance in the injectable lesions of melanoma patients.

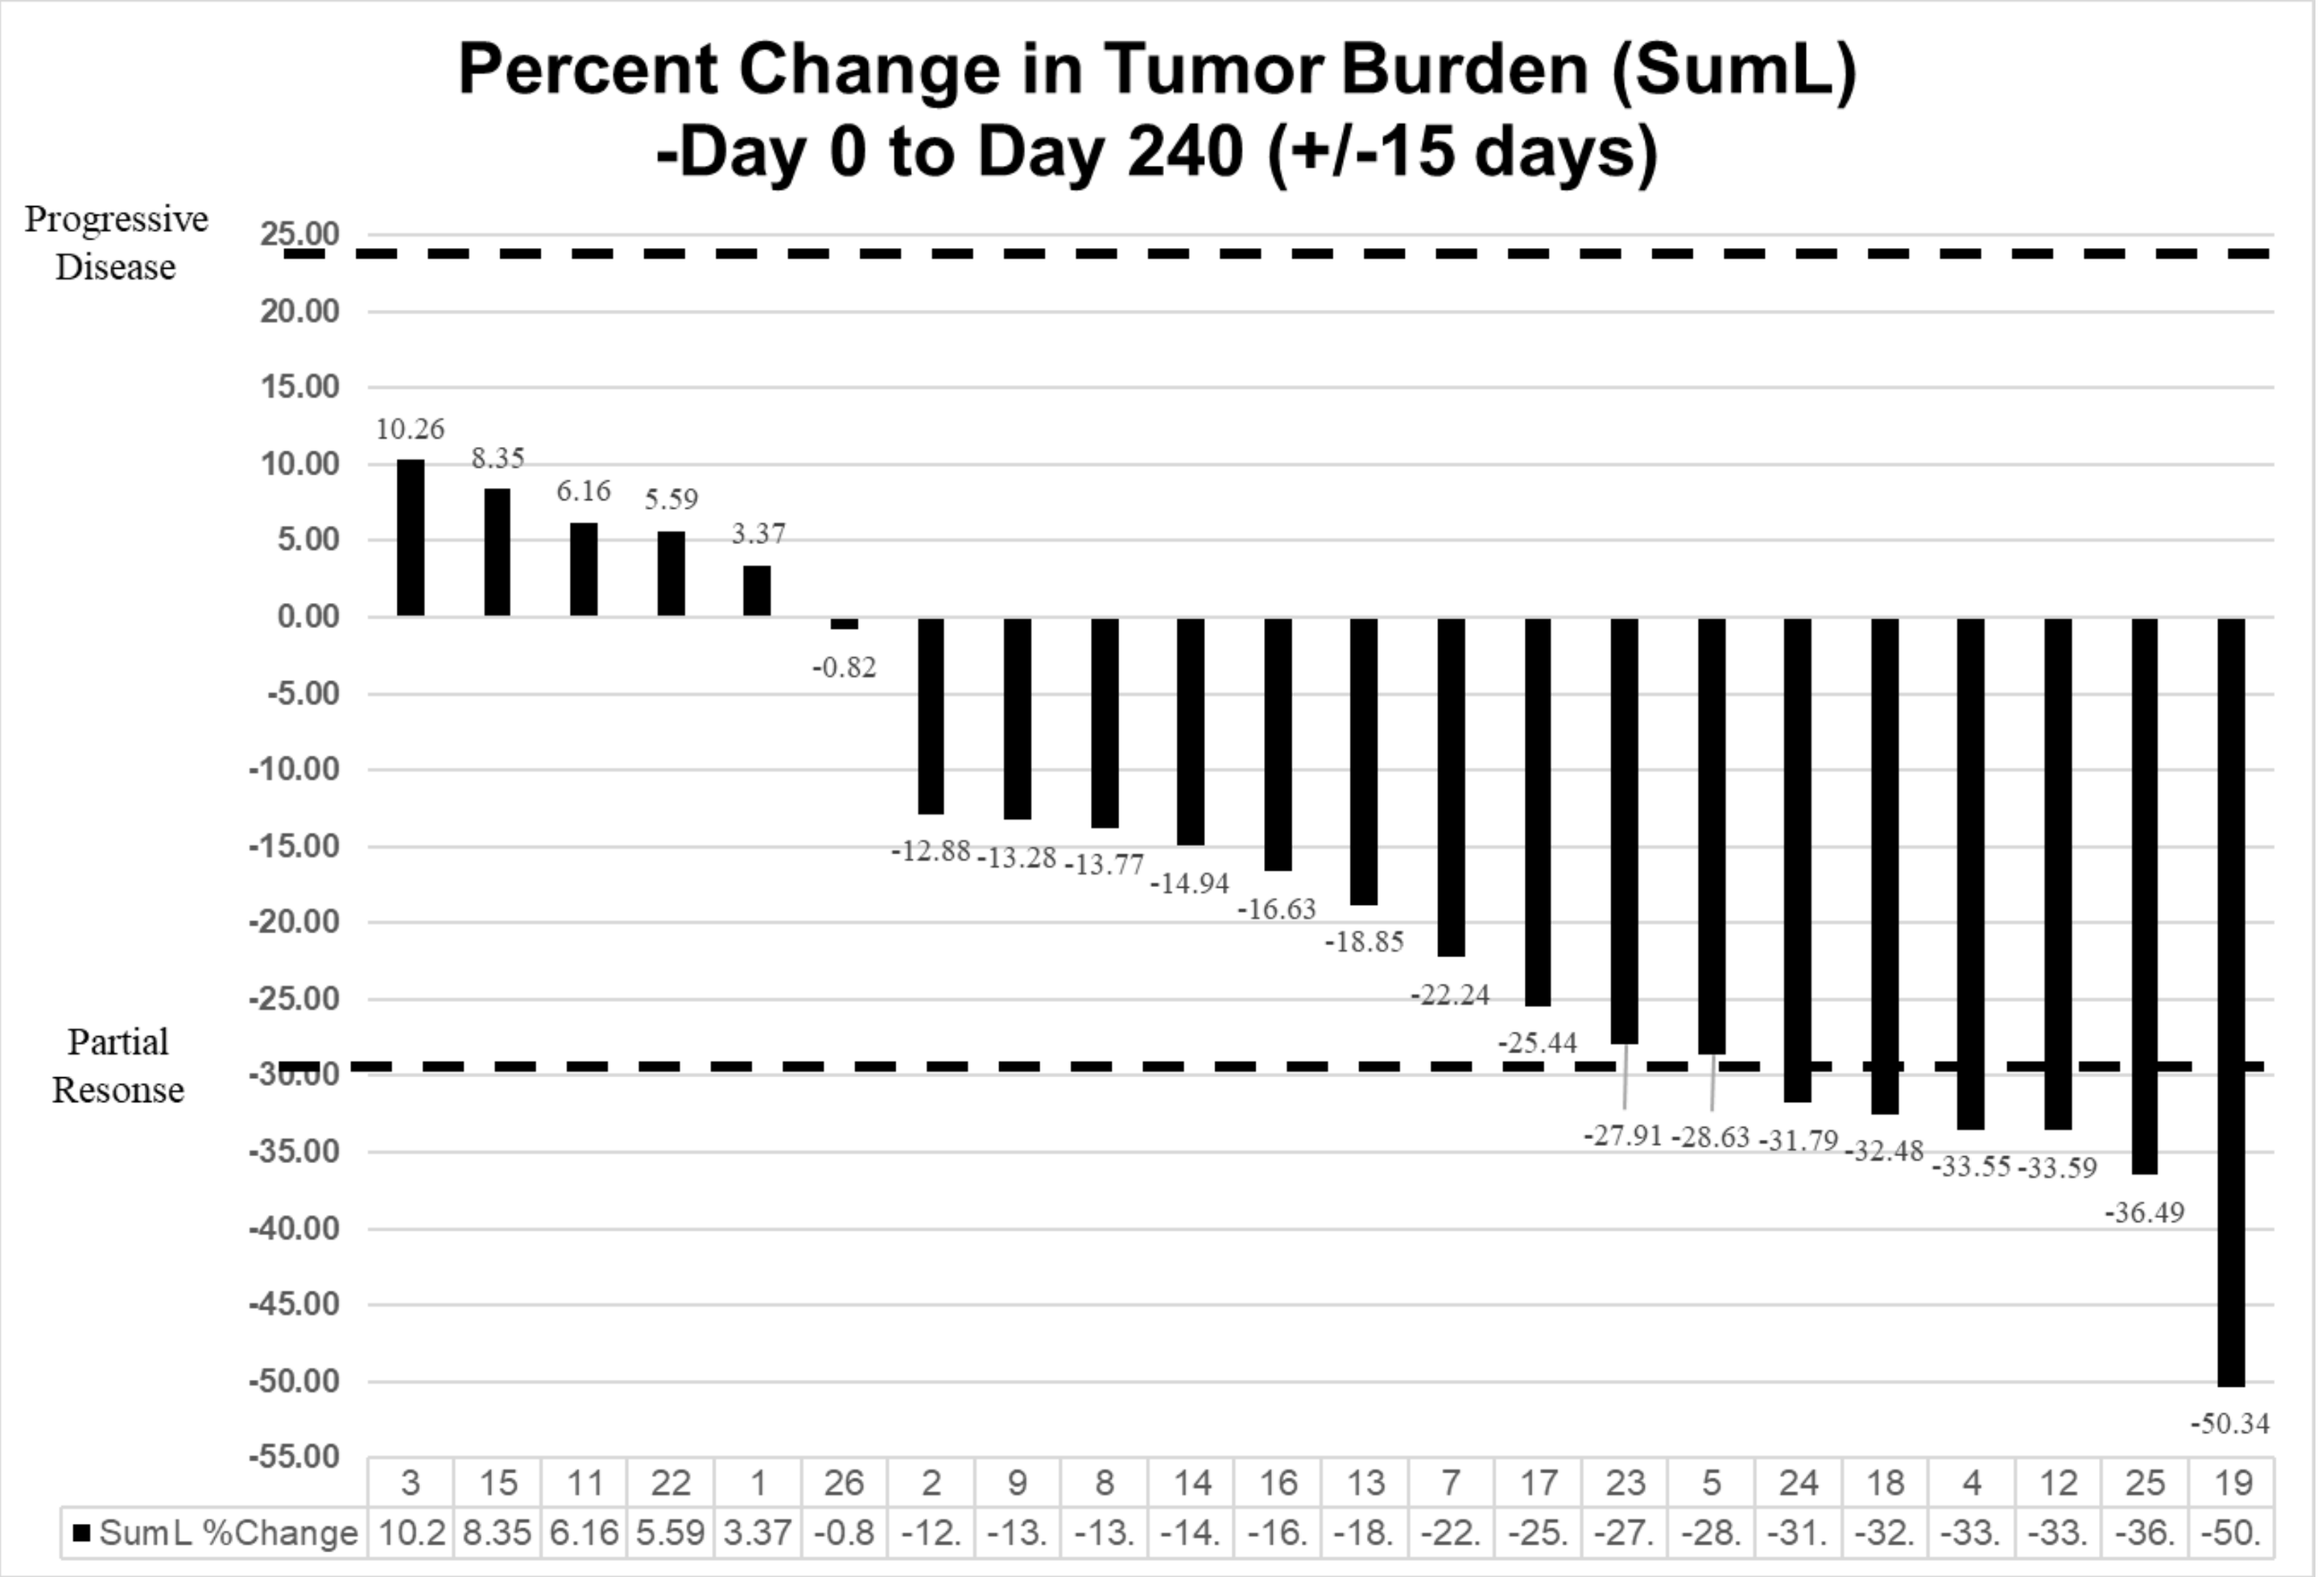

Percent Change in Tumor Burden (SumL)
-Day 0 to Day 240 (+/-15 days)
Progressive Disease
Partial Resonse
3 15 11 22 1 26 2 9 8 14 16 13 7 17 23 5 24 18 4 12 25 19
SumL %Change
10.26 8.35 6.16 5.59 3.37 -0.82 -12.88 -13.28 -13.77 -14.94 -16.63 -18.85 -22.24 -25.44 -27.91 -28.63 -31.79 -32.48 -33.55 -33.59 -36.49 -50.34

CLINICAL STUDY DESIGN

Approximately twenty adult patients (≥ 18 years of age), of any sex, ethnicity, and race with histologically confirmed MCC or cSCC with accessible lesions, will be eligible for study enrollment and treatment with IFx-Hu2.0 (i.e. 20 total patients across both indications). These types of non-melanoma cancers are very rare in the pediatric population (< 18 years of age) with only scattered case reports. The potential for development of this product for pediatric subjects with MCC or cSCC will be evaluated after the results of this study are available.

Patients must have at least one injectable lesion, defined as an easily palpable superficial lesion (cutaneous, subcutaneous or lymph nodal metastasis) that can be accurately localized, stabilized by palpation, and is superficial enough to enable intralesional injection.

To be eligible for this study, patients must have progressed despite standard therapy(ies), or are intolerant to or refused standard therapy(ies).

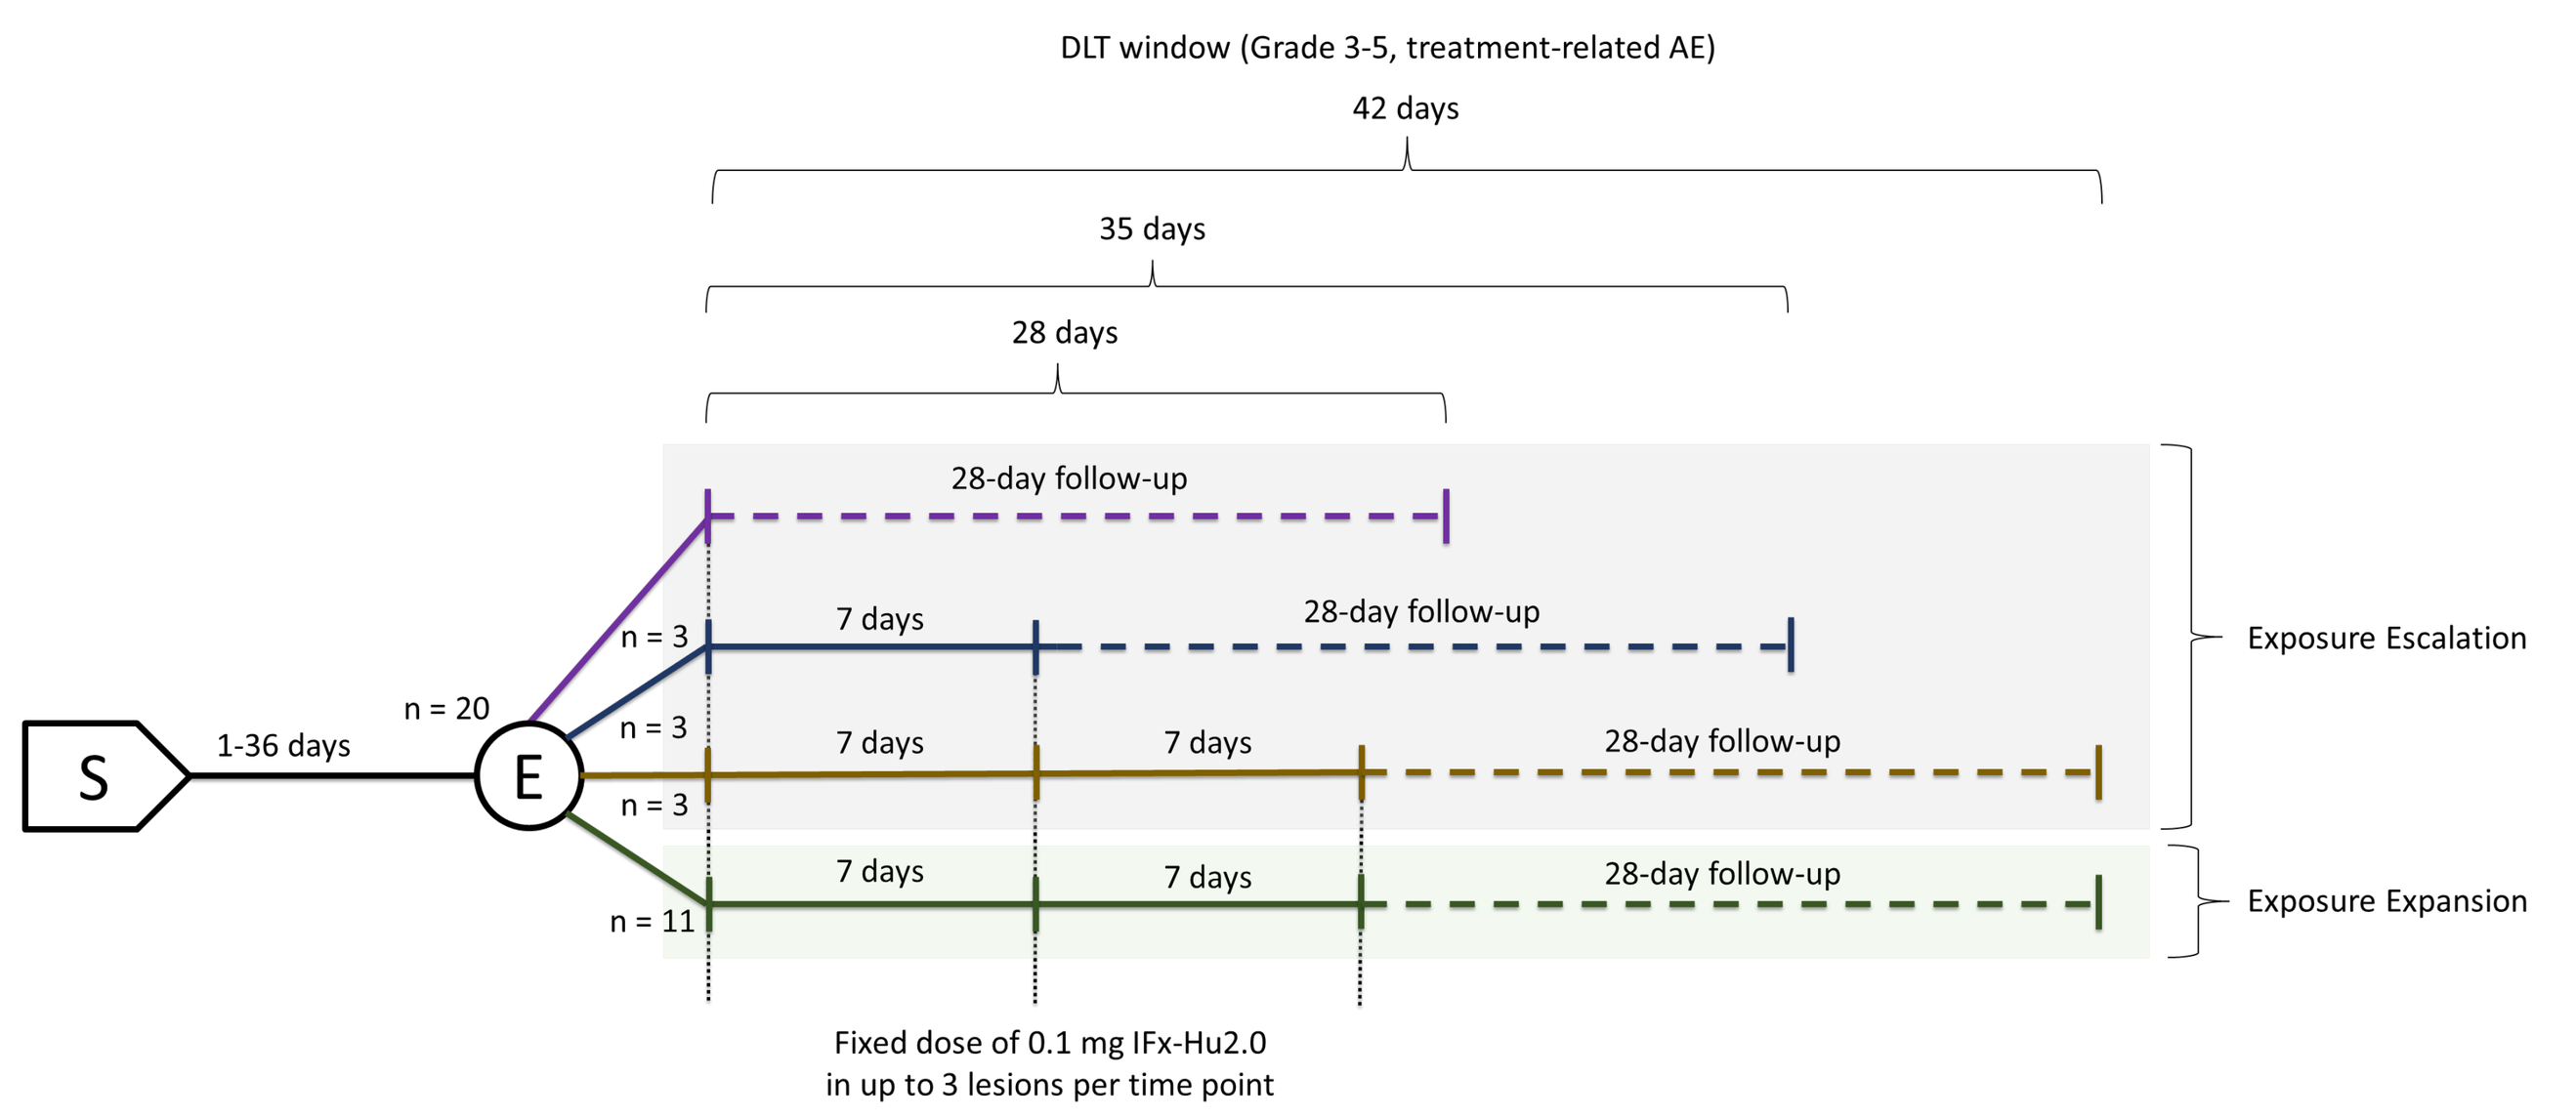

DLT window (Grade 3-5, treatment-related AE)
42 days
35 days
28 days
28-day follow-up
7 days
S
1-36 days
n = 20
E
n = 3
n = 3
n = 3
n = 11
Exposure Escalation
Exposure Expansion
Fixed dose of 0.1 mg IFx-Hu2.0
in up to 3 lesions per time point

REFERENCES

1. Boyle, M. D., J. Weber-Heynemann, R. Raeder and A. Podbielski (1995). "Characterization of a gene coding for a type IIo bacterial IgG-binding protein." Mol Immunol 32(9): 669-678.
2. Kim, T., R. N. Amaria, C. Spencer, A. Reuben, Z. A. Cooper and J. A. Wargo (2014). "Combining targeted therapy and immune checkpoint inhibitors in the treatment of metastatic melanoma." Cancer Biol Med 11(4): 237-246.
3. Ramiya VK, Jerald MM, Lawman PD, Lawman MJ. Autologous tumor cells engineered to express bacterial antigens. Methods Mol Biol. 2014;1139:243-57.
4. Morphogenesis USDA-CBER Field Safety and Efficacy Report; pAc/*emm55* in TE buffer as an aid in the treatment of melanoma in equines; January 2017.
5. Bunch B, et al., “Anti-tumor efficacy of plasmid encoding emm55 in a murine melanoma model” unpublished work.

ABOUT MORPHOGENESIS

Headquartered in Tampa, Florida, Morphogenesis is a cell and gene therapy company that is changing the way the immune system recognizes and eliminates tumor cells.

Our mission is to change the way chronic diseases are treated by engaging the innate intelligence of the body.