ASCENIV™ Reduces Viral Infections and Associated Comorbidities in a Primary Immunodeficiency Patient

Kevin P. Rosenbach, MD[1]; Benjamin N. Greener, PharmD, MS[2]; John T. Rosenbach[1]; Gene A. Wetzstein, PharmD, BCOP[3]
1. Naples Allergy Center, Naples, FL. 2. PPD, part of Thermo Fisher Scientific, Rockville, MD. 3. ADMA Biologics, Boca Raton, FL .

INTRODUCTION

- Primary immunodeficiency (PI) patients are at risk for recurrent severe bacterial and viral respiratory infections, even on immunoglobulin replacement therapy (IgRT) and prophylactic anti-infectives
- We report a challenging case of a PI patient who suffered from chronic and persistent infections on IgRT who was switched to ASCENIV (immune globulin intravenous [IGIV], human – slra, ADMA Biologics, Boca Raton, FL), a unique immune globulin produced from blending normal plasma and RSV hyperimmune plasma that contains elevated levels of antibodies to multiple respiratory pathogens

OBJECTIVE

- To describe the clinical use of ASCENIV in an adult with late-onset combined immune deficiency (LOCID) with a history of uncontrolled, recurrent respiratory infections

PATIENT CHARACTERISTICS

- 86-year-old male with a PI diagnosis of late-onset combined immune deficiency (LOCID) due to hypogammaglobulinemia in combination with impaired cellular immunity
- Past medical history included multiple uncontrolled recurrent bacterial and viral respiratory tract infections, lifelong allergies, nummular eczema, contact dermatitis, moderate persistent allergic asthma, bronchiectasis, and a former 40 pack/year smoking history
- The patient had required antibiotics, antivirals, and steroids at least every other month

CLINICAL COURSE/RESULTS

- The patient had been experiencing frequent recurrent upper respiratory infections (URIs) of both bacterial and viral origin that had resulted in bronchitis, with progressive pneumonia in some instances
- Workup at initial consultation revealed specific antibody deficiencies and impaired markers of cellular immunity
- The patient was started on subcutaneous immunoglobulin replacement therapy (SCIG) but URIs were not fully controlled and recurrent viral URIs continued, despite dose escalation and preventive antiviral therapies
 - IgG trough was maintained at >1600 mg/dL
- The patient was switched to ASCENIV, a commercially available immunoglobulin product that is produced from blending normal and RSV hyperimmune plasma, resulting in an Ig with a unique antibody profile with elevated levels of titers against clinically concerning respiratory viruses
- Following initiation of ASCENIV, the patient demonstrated significant clinical improvement and continues to report that the persistent URIs have resolved with no further infections and a reduction in anti-infective utilization
- The patient reports improved asthma and symptomatic control and no longer requires systemic corticosteroids, nebulizer therapies, metered dose inhalers, or symptomatic therapies to control mucus and cough
- ASCENIV has been well tolerated

CONCLUSIONS

- This case report demonstrates the clinical improvements of a challenging LOCID patient after being switched to ASCENIV from standard IgRT
- Patient continues to be managed on ASCENIV therapy
- ASCENIV may provide a substantial clinical benefit in immunodeficient patients who continue to experience breakthrough infections despite administration of standard IgRT, or in high-risk patients that possess both humoral and cellular immunity defects