Staphyloccocal Protein A, Which Is Immuno-Modulatory In The Murine CIA Model, Does Not Increase Infection Severity In Murine Listeria Or Candida Challenge Models, In Contrast To Anti-TNF Treatment

Edward Bernton[1] and Valerie Lowe,[2] [1]Protalex Inc., [2]Washington Biotechnology, Inc.

ABSTRACT

Background: PRTX-100 is a highly-purified GMP staphylococcal protein A (SpA) that is currently in Phase I trials in patients with active rheumatoid arthritis (RA). SpA has diverse activities in vitro and in vivo: forming immune complexes with IgG, SpA induces a "suppressor" phenotype in murine and human macrophages; IP or IV administration reduces disease severity in the murine CIA model. SpA can also inhibit activation of human monocyte-derived macrophages by LPS and gamma-IFN. SpA binds to Vh3 B-lymphocytes and relocates with them to lymphoid tissues. Since anti-cytokine biologic DMARDS, in particular anti-TNF products, have been shown to increase patient susceptibility to pathogens such as listeria, fungi, and tuberculosis, we compared the effects of SpA treatment to that of etanercept and anti-mouse TNF in murine models of Listeria and Candida infection.

Methods: For Listeria challenges, groups of 15 Balb/C mice were treated IP with either 10 mL/kg of 0.1% BSA, 15 mg/kg of etanercept, 50 or 250 µg/kg of SpA, or 0.2 mL of rabbit anti-mouse TNF antisera. After 4 hours mice were administered 5 x 10^{10} CFU of *L. monocytogenes* orally. Mice were then re-treated with drugs every 48 hours x 2. Weights and mortality were recorded daily. On Days 3, 5, and 8, five mice/group were sacrificed for spleen cultures and CFU counts. For Candida challenges, groups of 15 female CD-1 mice received the same treatments. Four hours after the first treatments mice were injected IV with 2 x 10^6 CFU of *Candida albicans*. Daily weights and mortality were recorded. On Days 3, 5, and 8, five mice/group were sacrificed for kidney cultures and CFU counts.

Results: Mean values for weights, bacterial load: *Listeria challenge – BSA*: 20% mortality.10% weight loss at Day 4 was regained by Day 6. *Anti-TNF*: 21% weight loss on Day 5, 100% lethality by Day 6. *Etanercept*: 17% weight loss by Day 5, maintained through to end of study; no mortality. Spleen bacterial counts were higher (p<0.05) than with BSA treatment at Day 5. SpA, 25 or 250 µg/kg; no mortality, weight loss similar to BSA-treated mice. Lower bacterial counts at Day 5 than seen following etanercept or anti-TNF treatment. *Candida challenge – BSA*: 17% weight loss by Day 5 and 10% mortality by Day 8. *Anti-TNF*: 23% weight loss on Day 5 with 100% mortality by Day 6. Kidney yeast counts higher (p<0.05) than BSA group on days 3 and 5. *Etanercept*: 30% weight loss by Day 7 and 60% mortality by Day 8. Mean kidney counts higher (p<0.05) than BSA group at days 3, 5, and 8. *SpA groups*: weight loss similar to BSA group. Kidney counts not significantly different at days 5 and 8 from BSA group; no mortality by Day 8.

Conclusion: In contrast to etanercept or anti-TNF treatment, repeated injections of SpA at 50 or 250 µg/kg did not affect disease severity or pathogen load in these challenge models with bacterial or fungal intracellular pathogens.

CONTACT

Edward Bernton M.D.
Protalex Inc.
Email: ebernton@protalex.com
Phone: 240 920 8571
Website: www.protalex.com

Poster Template Designed by Genigraphics ©2012
1.800.790.4001 www.genigraphics.com

INTRODUCTION

- **PRTX-100** is a highly-purified staphylococcal protein A (SpA) with immuno-modulatory activities, which is in clinical development for the treatment of rheumatoid arthritis.
- Other biologic disease modifiers used for RA treatment, such as anti-TNF therapeutics, are labeled with "black-box" warnings describing an increased incidence of certain intracellular infections such as listeria and tuberculosis, and increased risk of systemic fungal infections.
- As a therapeutic without anti-cytokine activities, we investigated whether SpA would increase severity or lethality of infections in mice challenged with listeria or candida, using well-established models.
- Etanercept and rabbit-anti-mouse TNF-alpha, both of which are effective in the murine CIA model, were used as comparators.

METHODS AND MATERIALS

Table 1. Treatment Groups

Group	Treatment	Dose
1	0.1% BSA vehicle	10/mL/kg
2	Etanercept	15 mg/kg
3	SpA (PRTX-100)	50 mcg/kg
4	SpA (PRTX-100)	150 mcg/kg
5	*Anti-mouse TNF-alpha	0.15 mL/mouse

*High-titer rabbit anti-sera was precipitated with MES buffer, then dialyzed against PBS to give a semi-purified Ab fraction with 4–8 mg/mL of IgG.

Listeria challenge

- Day 1: fasted mice injected IP with treatment, and 4 hours later administered 5 x 10^{10} CFU of *L. monocytogenes*, PO, then re-fed. Mice were weighed and observed daily on Days 1–8.
- Day 3: five mice/group were randomly selected, sacrificed, and spleens frozen for bacterial culture. Remaining mice injected IP with study treatment.
- Day 5: five mice/group were sacrificed for spleen cultures. Remainder were injected IP with study treatment.
- Day 8: remaining mice were weighed, then sacrificed for spleen cultures.
- Endpoints: change in weight (by day), survival, spleen CFU Listeria. There were 15 mice per treatment group.
- Experiment was repeated without the etanercept treatment group, using the same treatment schedule, with 30 mice per group (7 sacrificed on Day 4, and 8 on Day 6 for spleen cultures) Remaining mice observed for weight loss and mortality until day 10.

Candida Challenge

- Day 1: mice injected IP with treatment, and 4 hours later injected IV with 2x10^6 *C. albicans*.
- Mice were weighed and observed daily on Days 1–8.
- Day 3: five mice per group were randomly selected, sacrificed, and kidneys frozen for fungal cultures. Remaining mice injected IP with study treatment.
- Day 5: five mice per group sacrificed for kidney cultures. Remaining mice injected IP with study treatment.
- Day 8: remaining mice weighed, then sacrificed for kidney cultures.
- Endpoints: change in weight (by day), survival, spleen CFU Listeria.
- There were 15 mice per treatment group.
- Experiment was repeated with 30 mice per group without etanercept (N=15). Same treatment schedule as before. Seven mice sacrificed for kidney cultures on Day 4 and eight sacrificed on Day 7. Remaining mice observed for weight loss and mortality until Day 9.

RESULTS

Listeria challenge: Survival

Listeria – Experiment 1

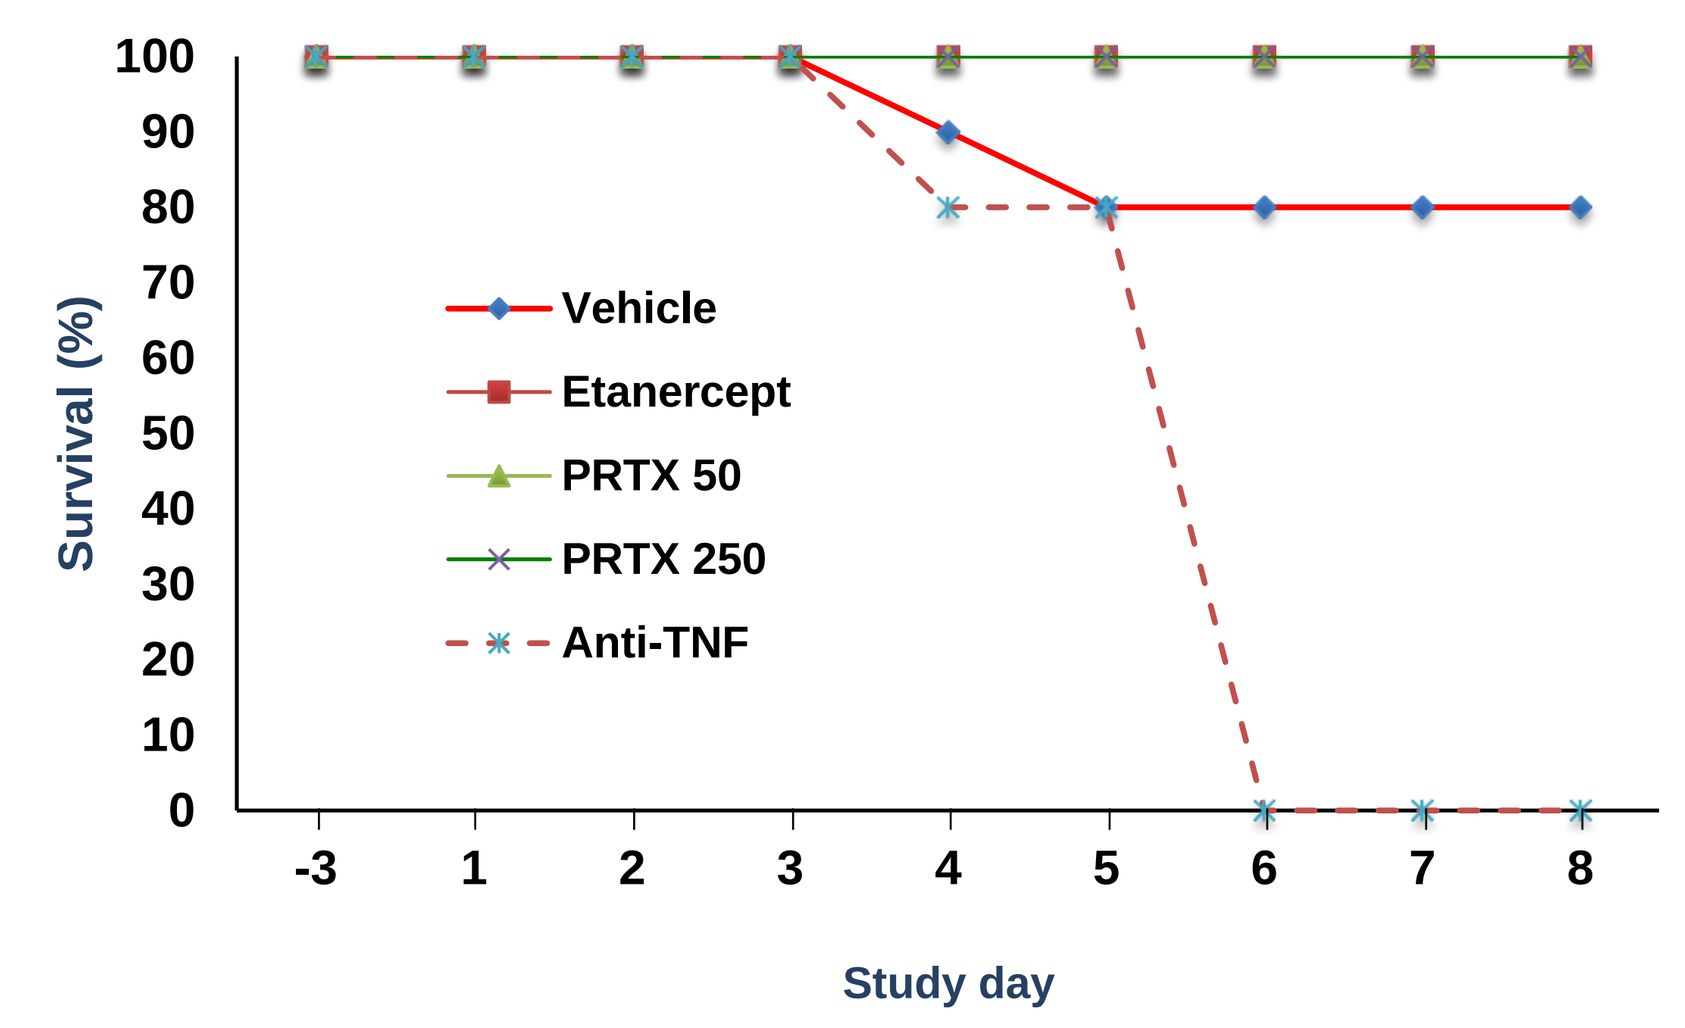
Survival (%)
Vehicle
Etanercept
PRTX 50
PRTX 250
Anti-TNF
Study day

Listeria – Experiment 2

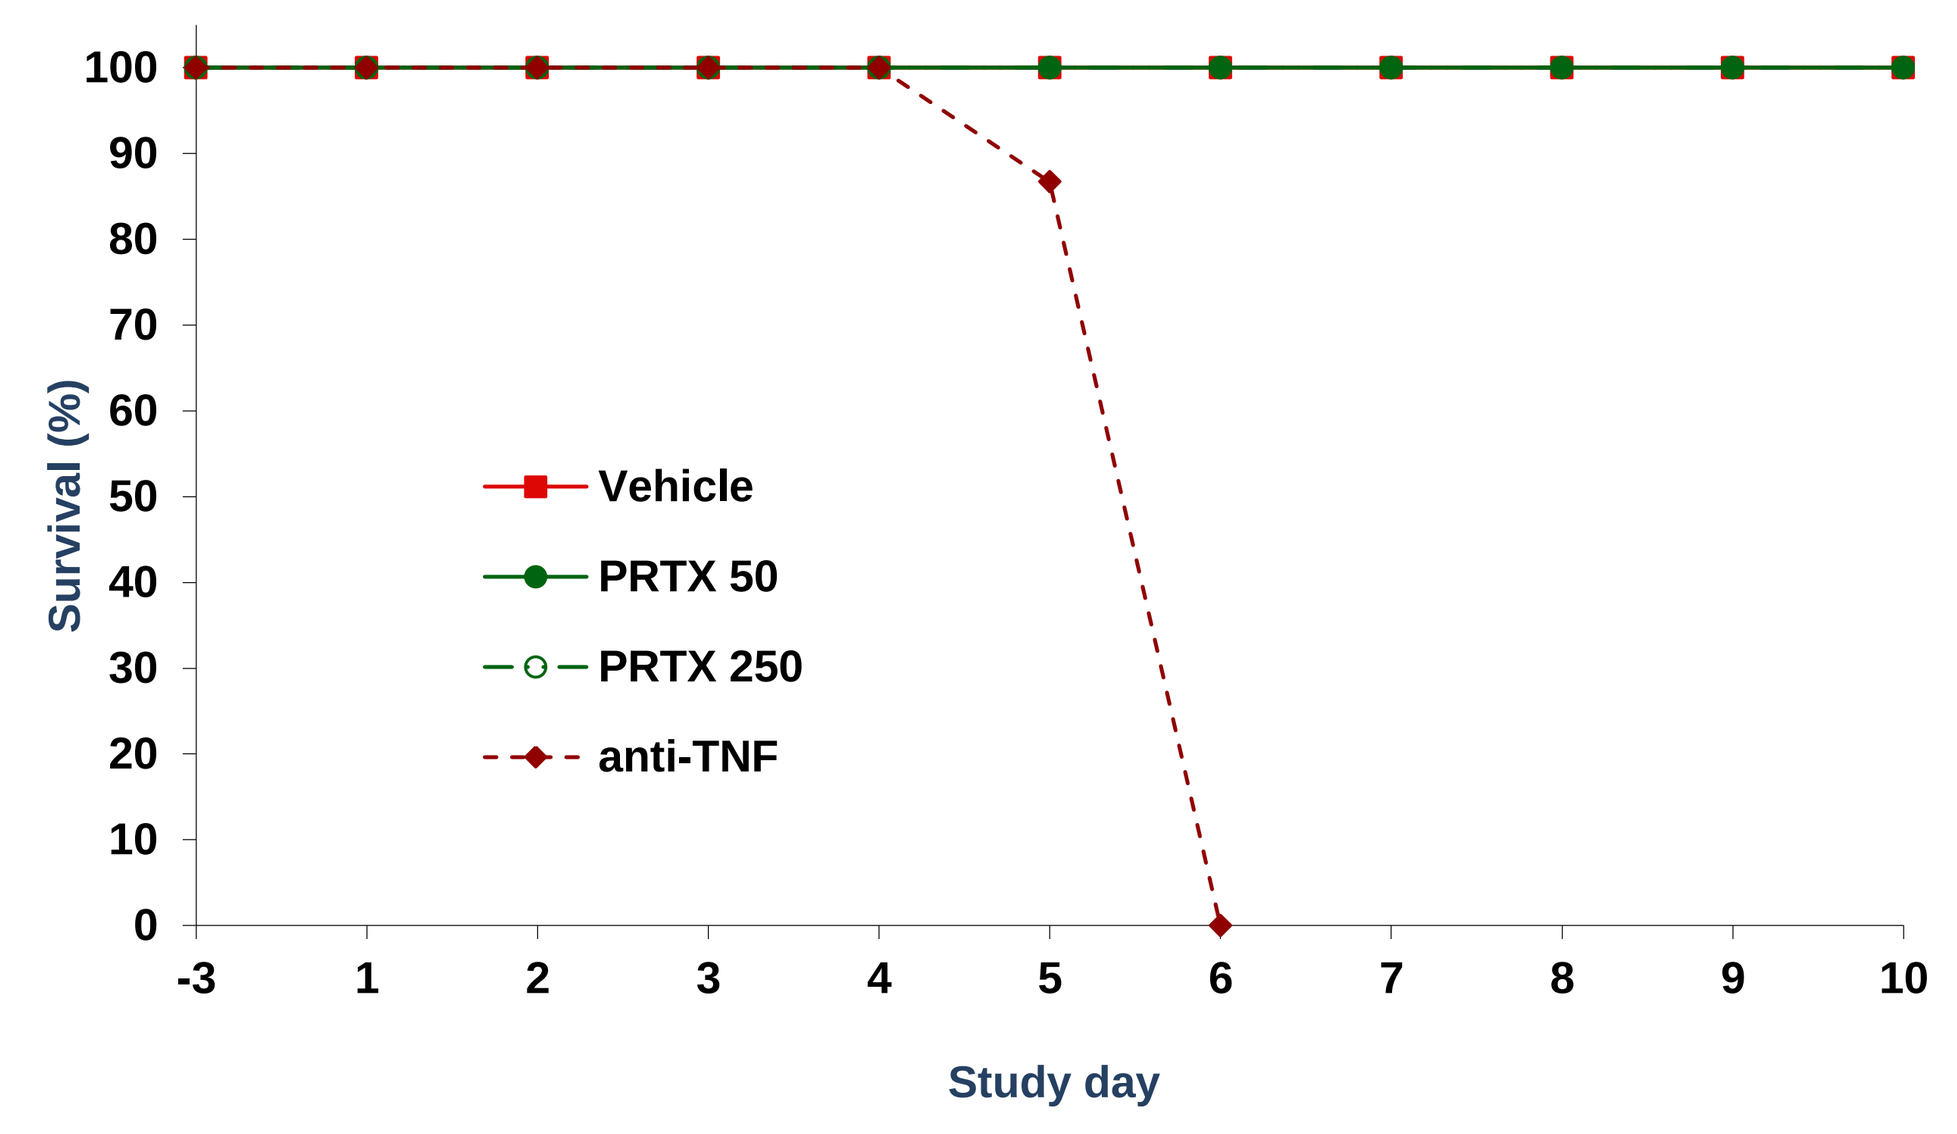
Survival (%)
Vehicle
PRTX 50
PRTX 250
anti-TNF
Study day

Listeria challenge: weight loss, bacterial load

Listeria Experiment 1: Effect of study treatment on diseased mouse weight. Etanercept and anti-TNF mean weights less than vehicle group (p<0.05 Day 6 to end). (Treatment of <u>non-infected</u> mice with anti-TNF caused no weight loss or mortality).

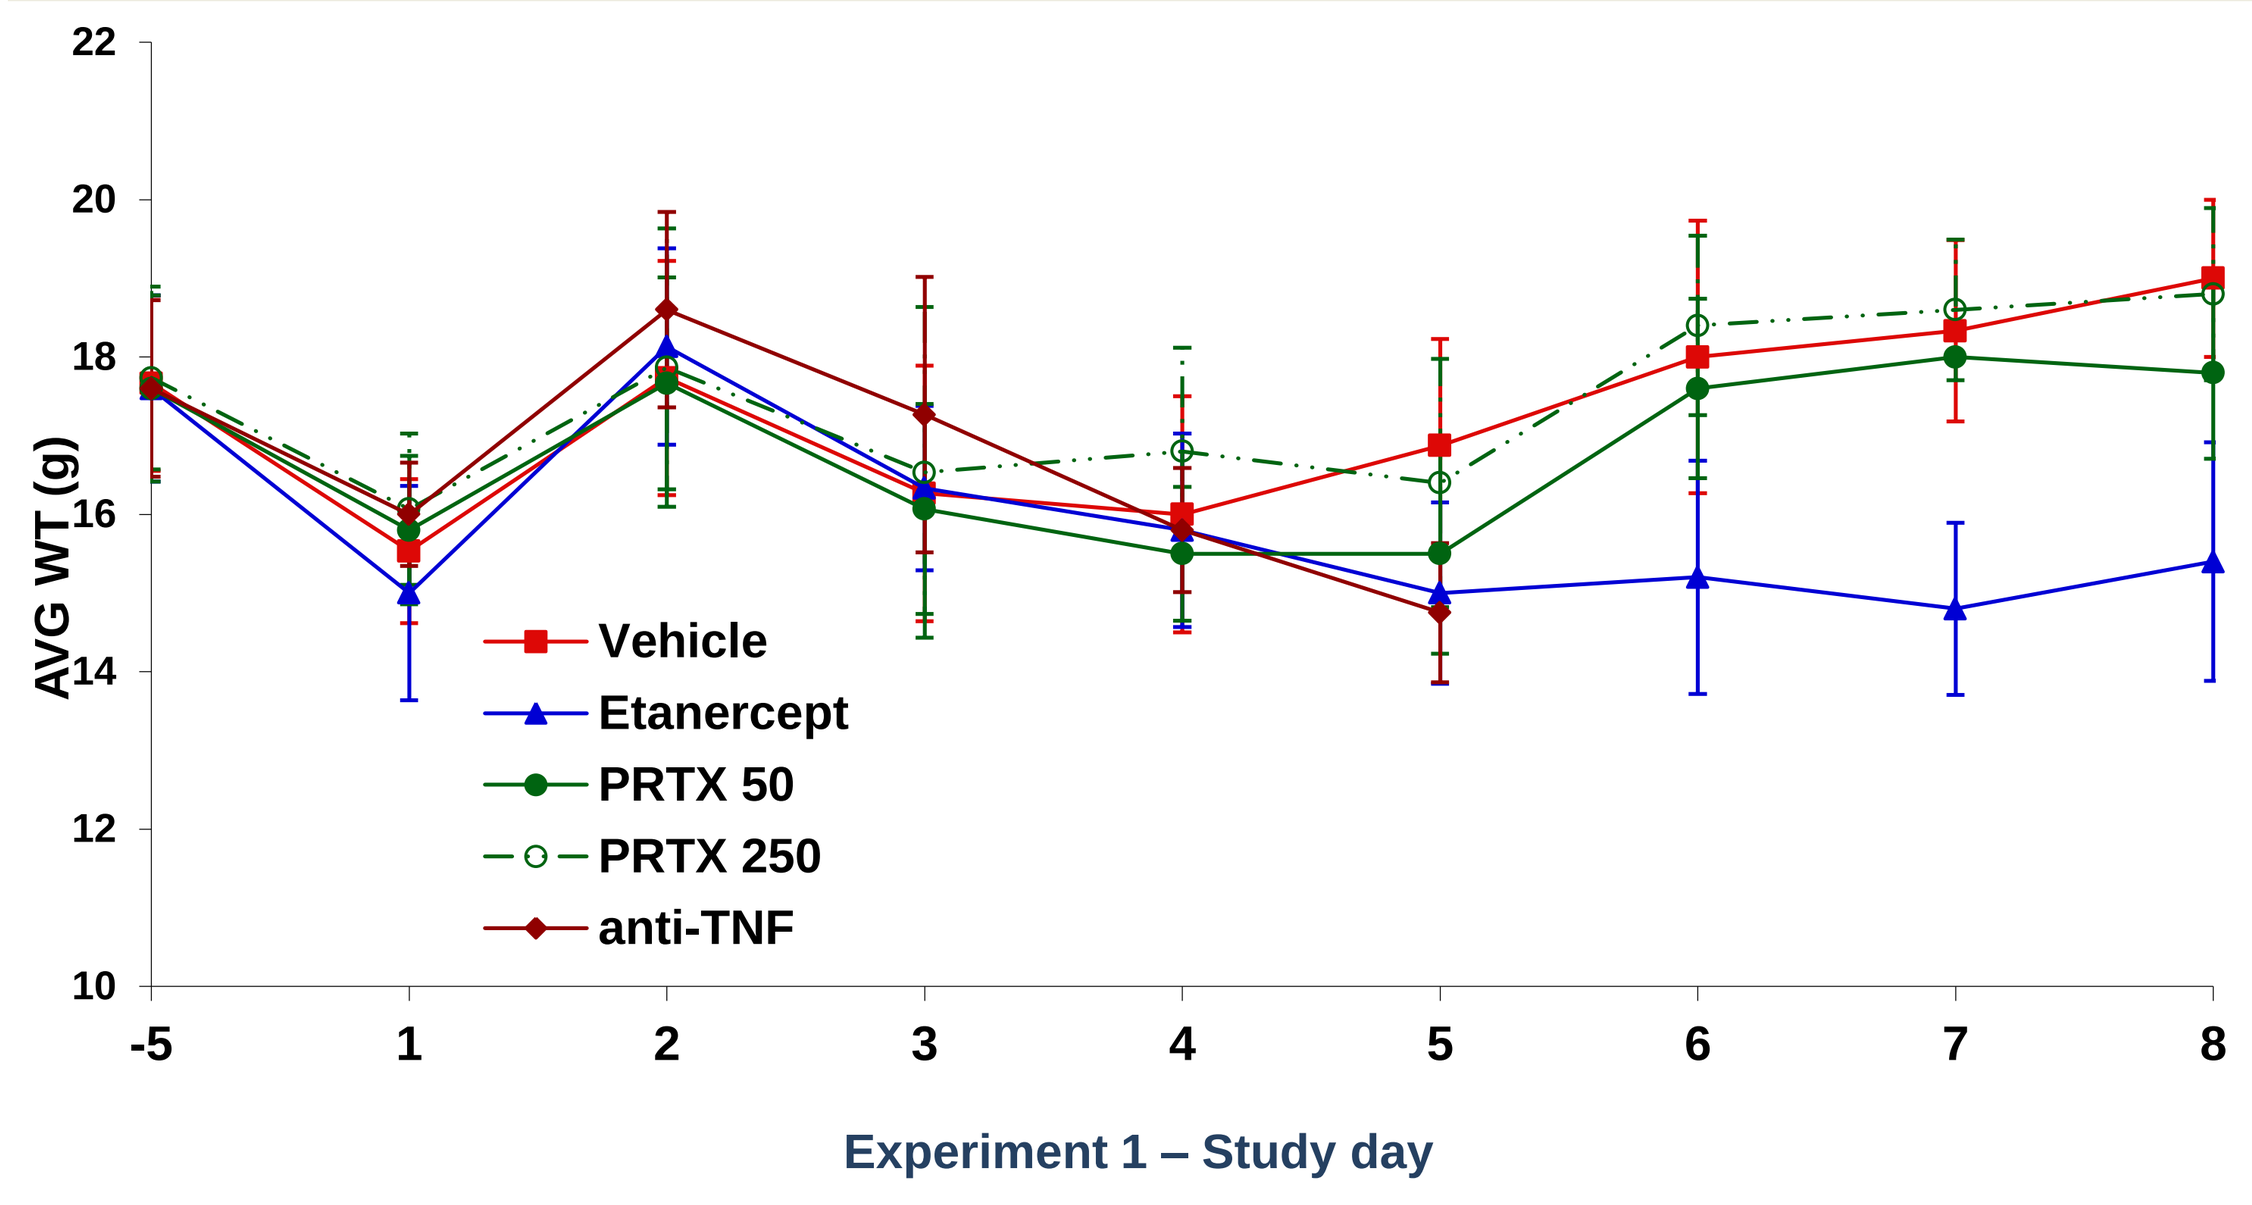
AVG WT (g)
Vehicle
Etanercept
PRTX 50
PRTX 250
anti-TNF
Experiment 1 – Study day

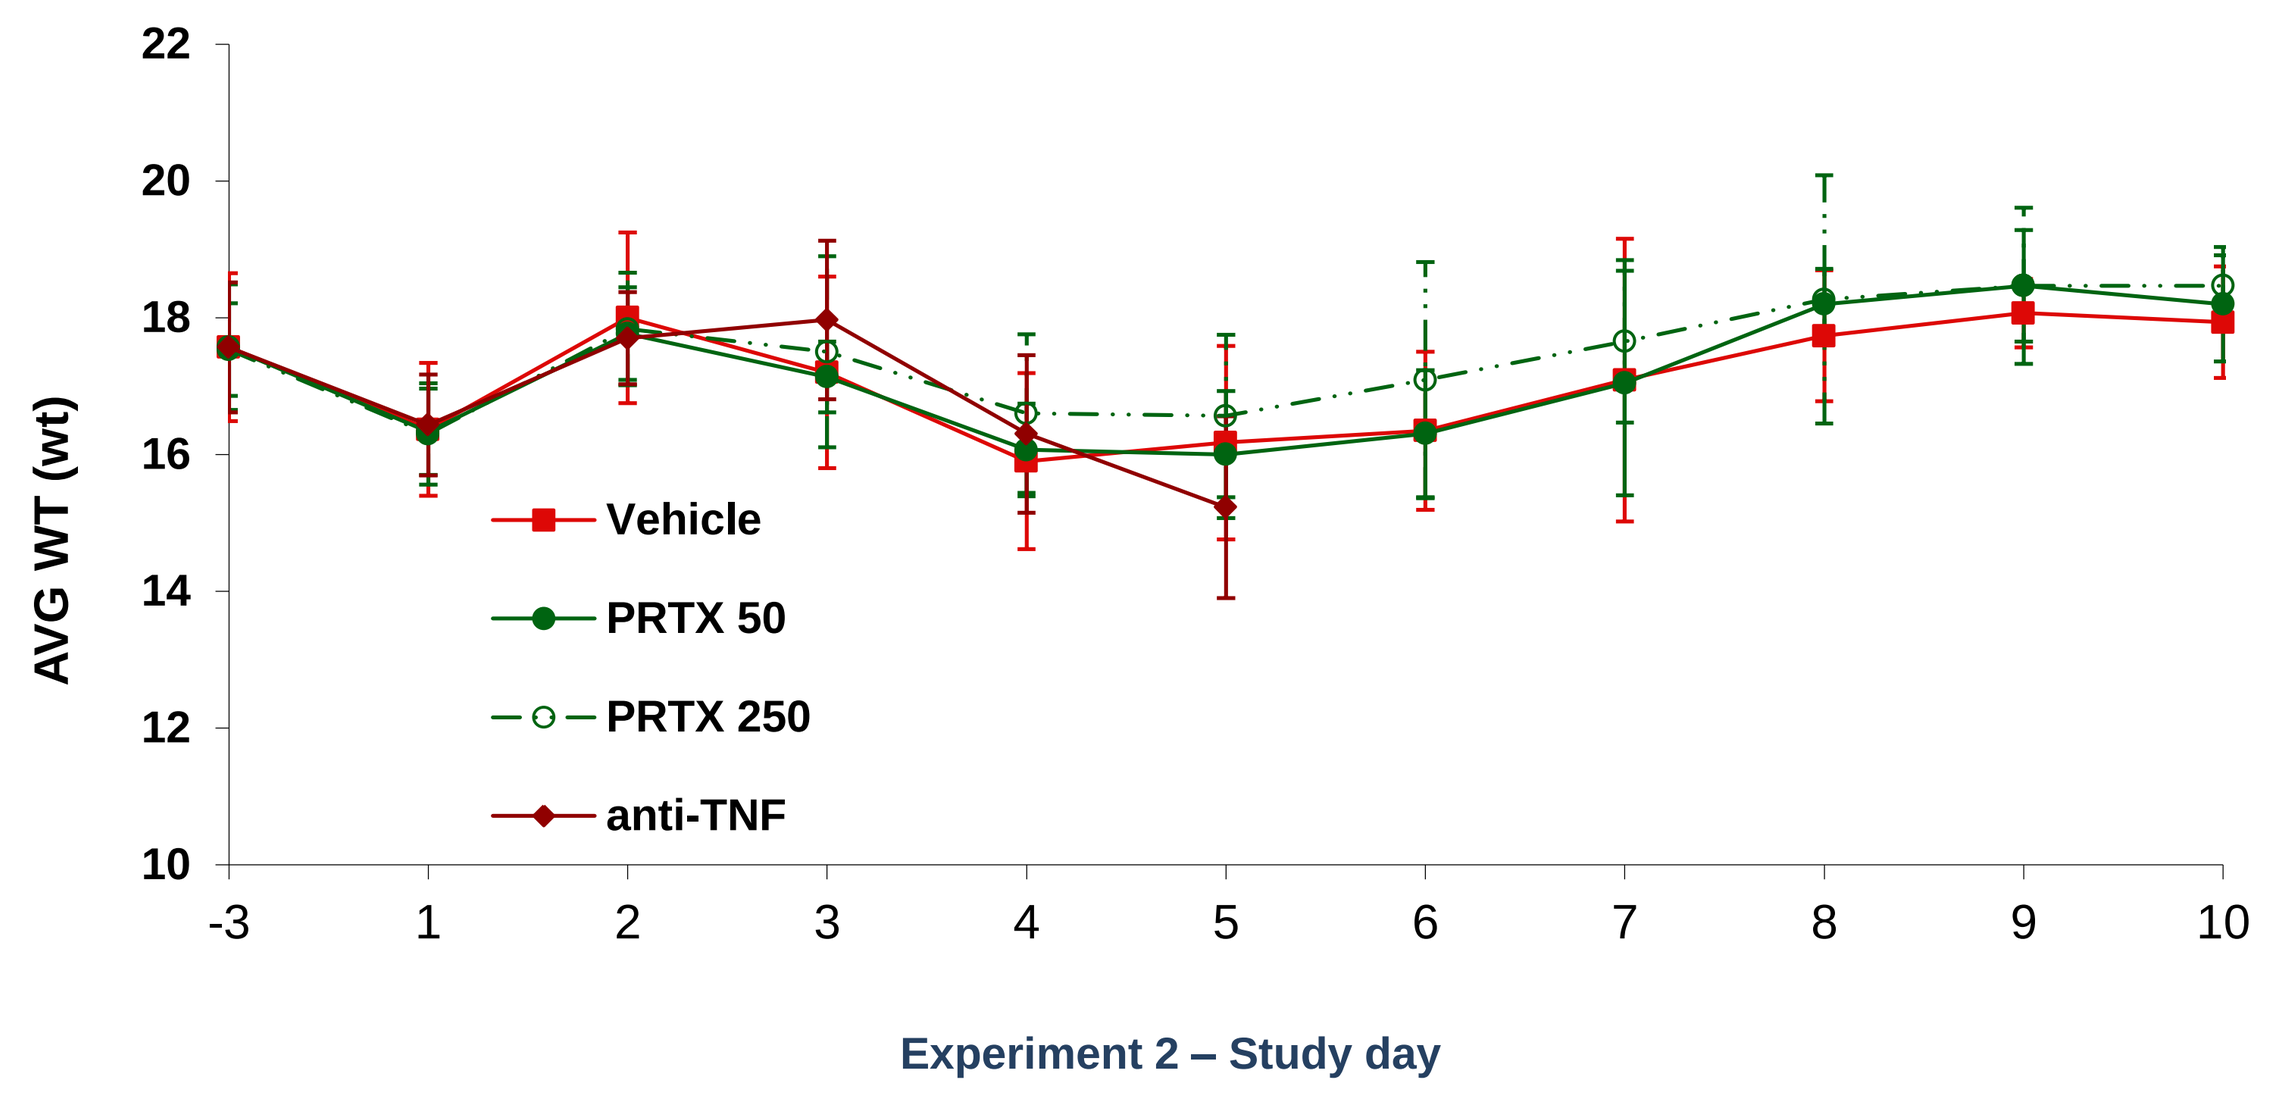
AVG WT (wt)
Vehicle
PRTX 50
PRTX 250
anti-TNF
Experiment 2 – Study day

Listeria – log of CFUs per spleen – Experiment 2

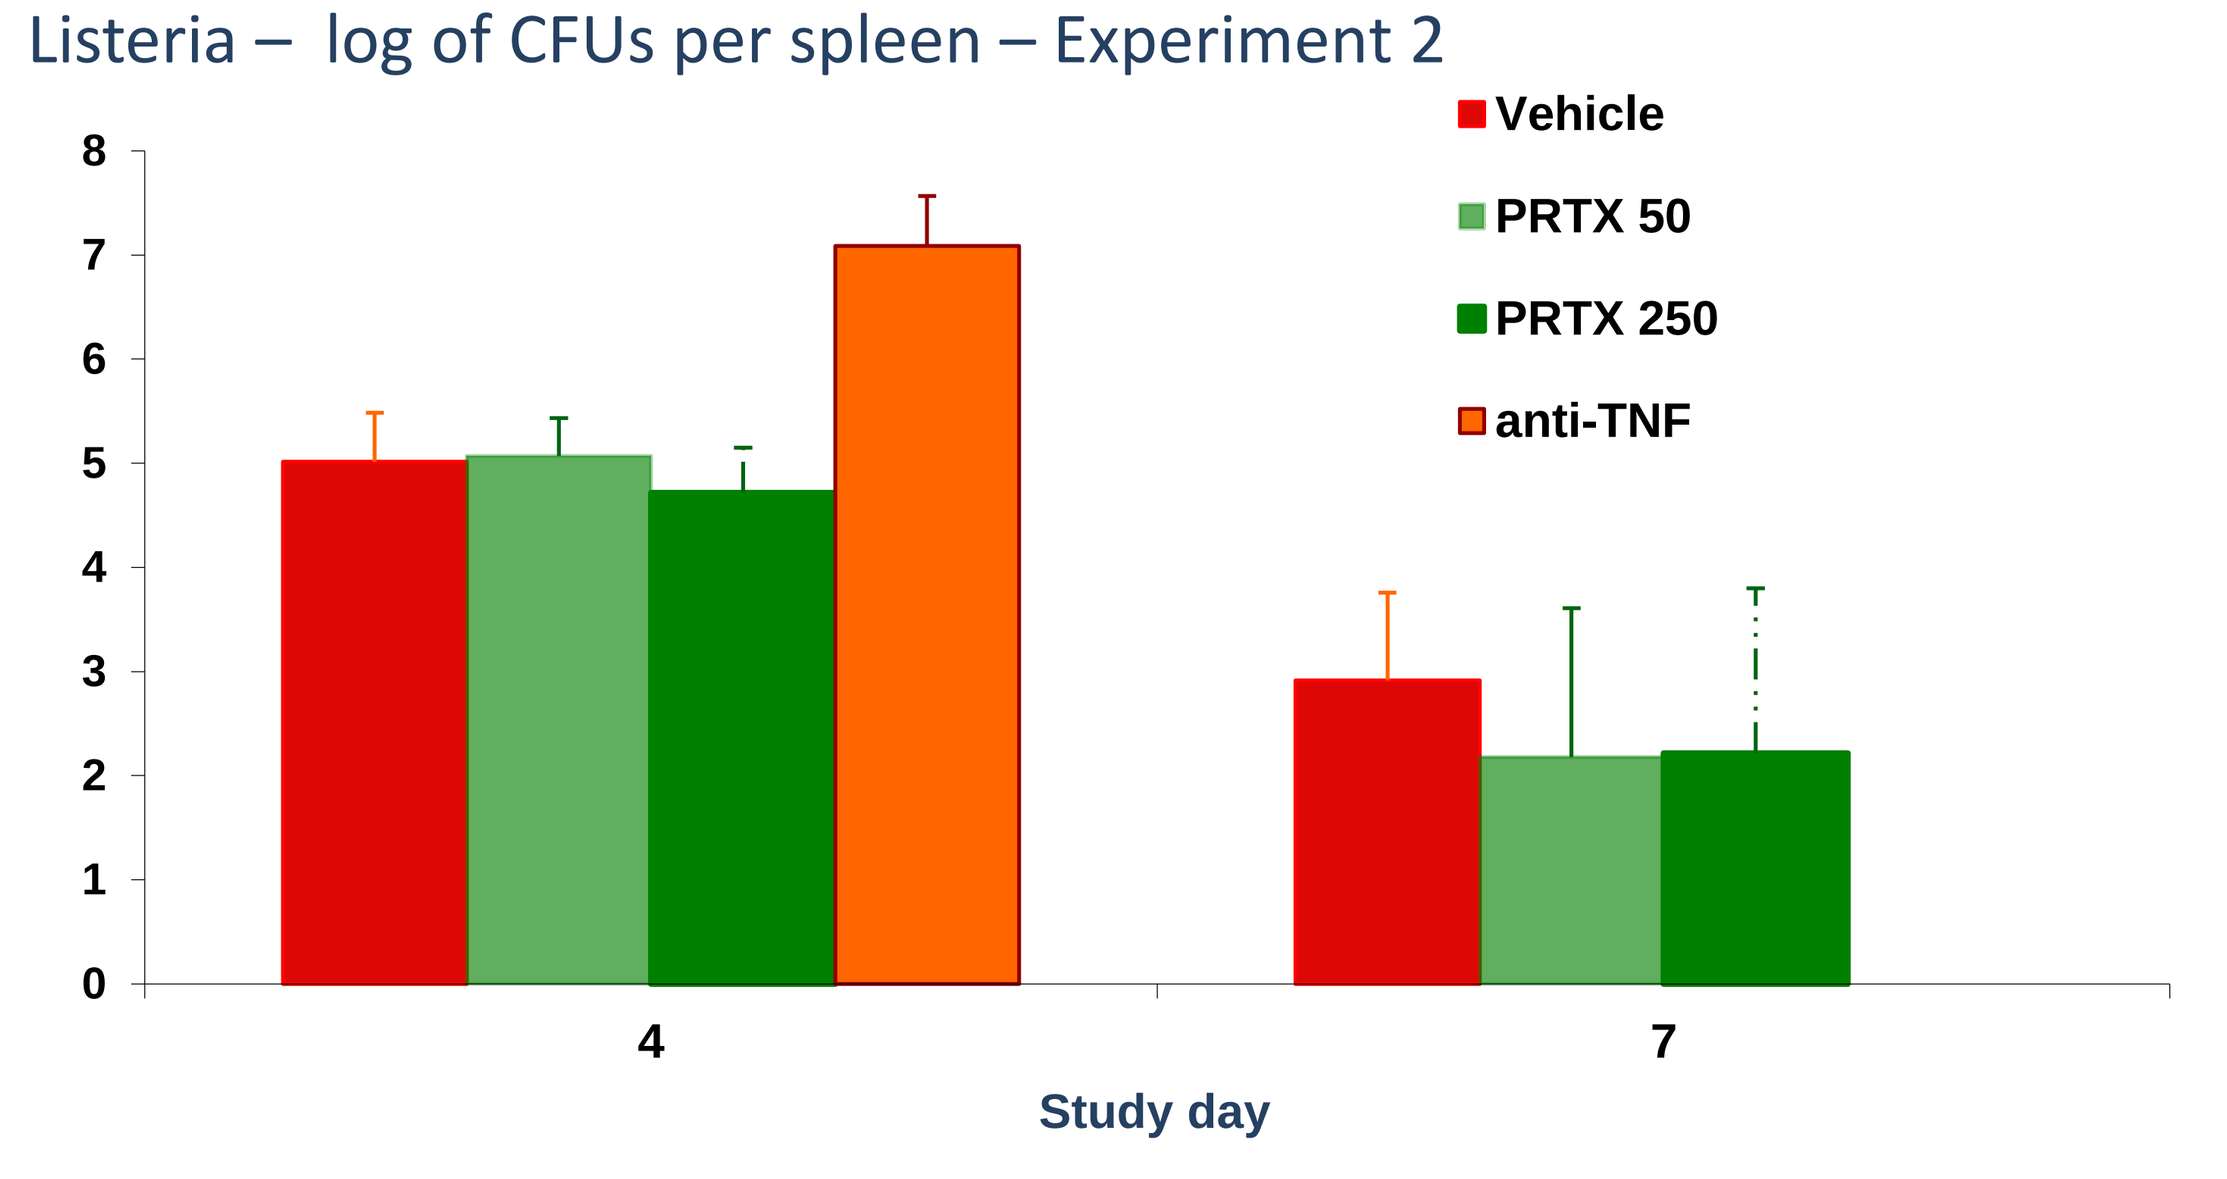
Vehicle
PRTX 50
PRTX 250
anti-TNF
Study day

Candida challenge: Survival, Kidney Fungal Load

Candida experiment 1- Survival : vehicle 80%, etanercept 40%, PRTX 50 & 250 100%, anti-TNF 0%**. Day 5 kidney CFUs**: etanercept and anti-TNF treatment groups higher than vehicle or PRTX 50 or PRTX 100 treatment groups (p<0.05).

Candida experiment 2 - kidney cultures not performed for etanercept group

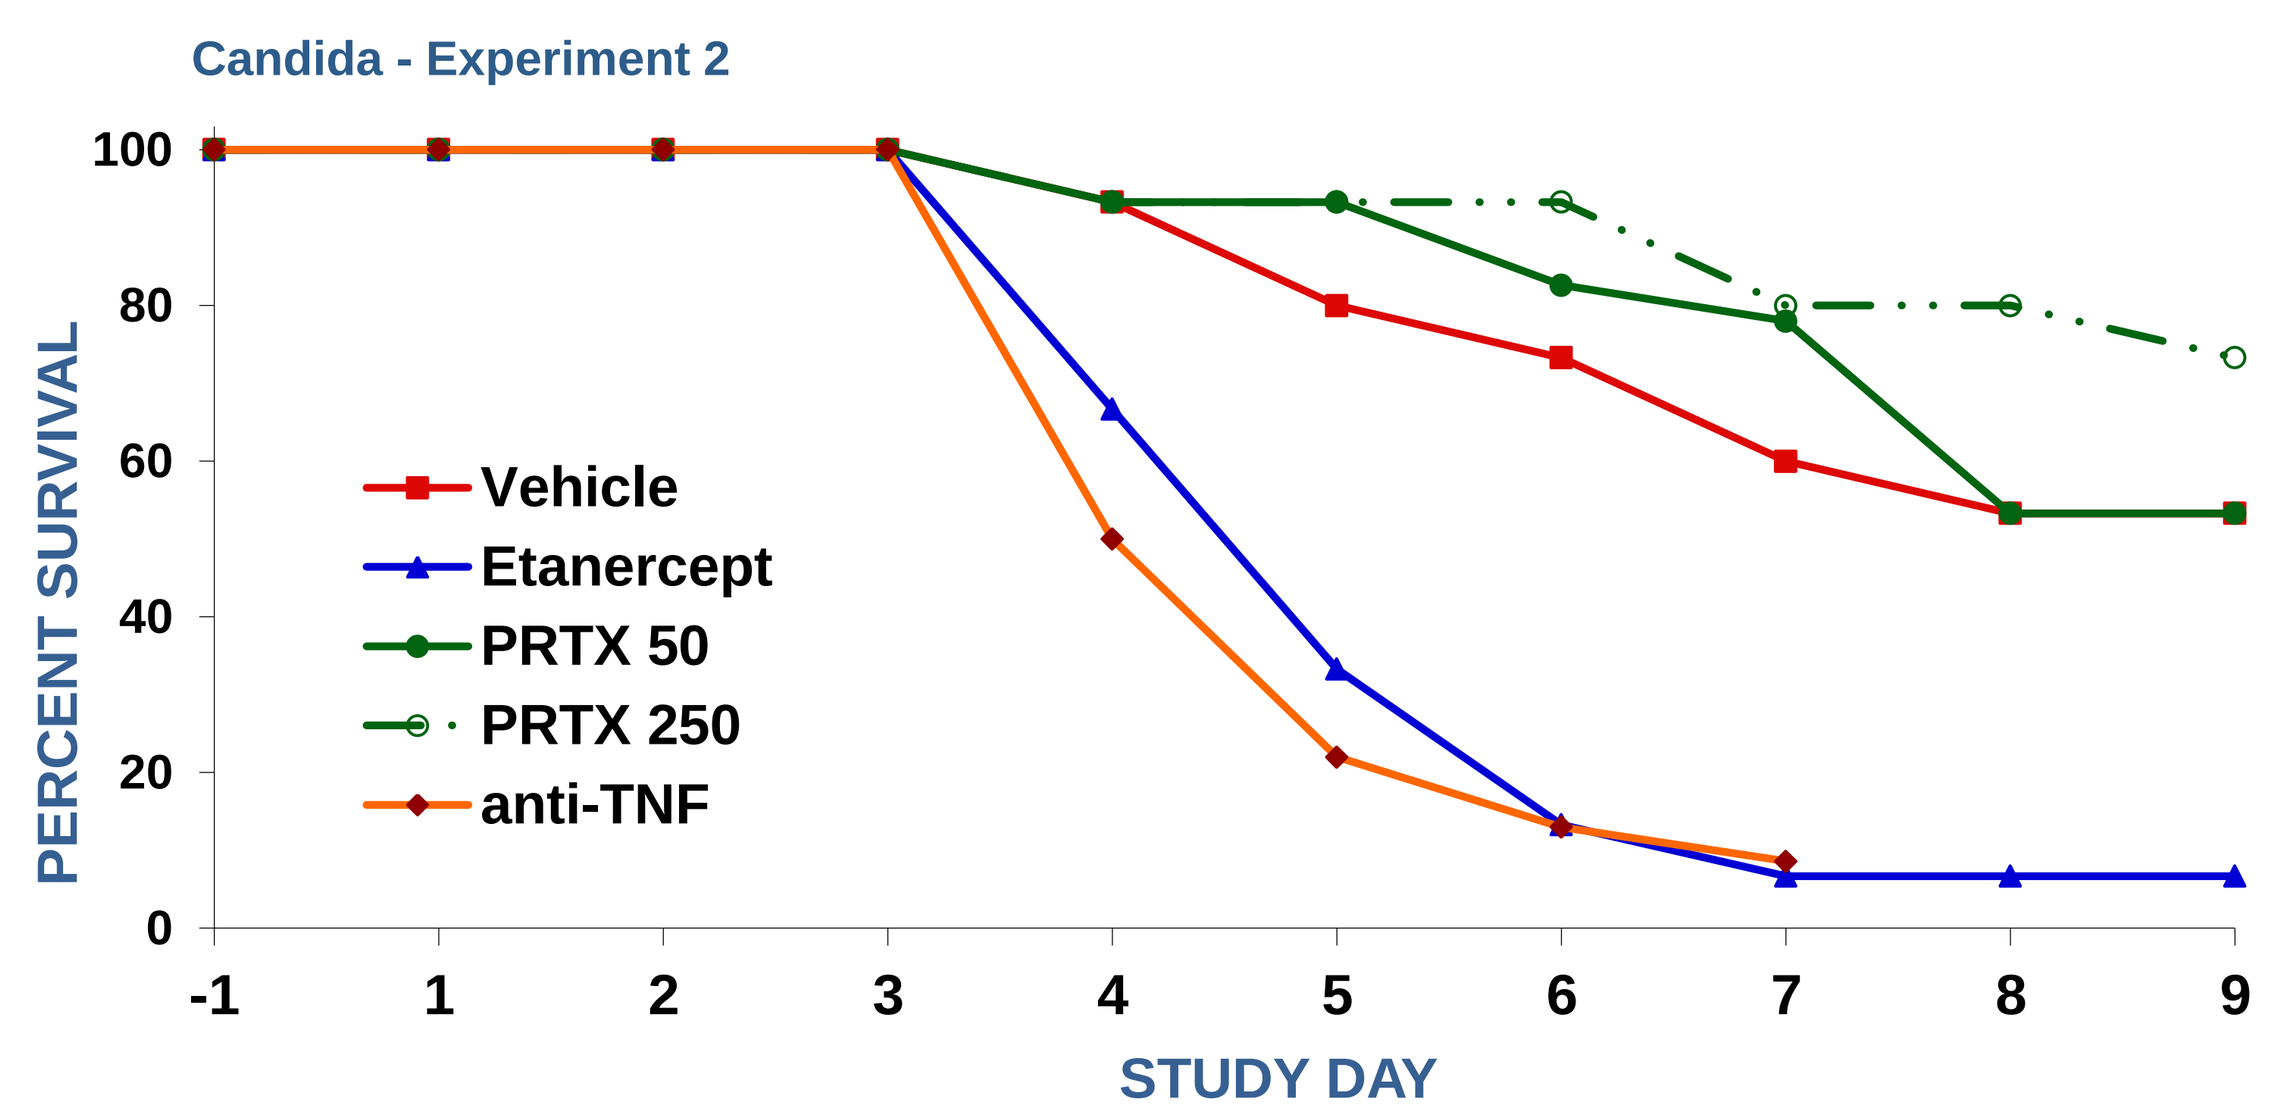
Candida - Experiment 2
PERCENT SURVIVAL
Vehicle
Etanercept
PRTX 50
PRTX 250
anti-TNF
STUDY DAY

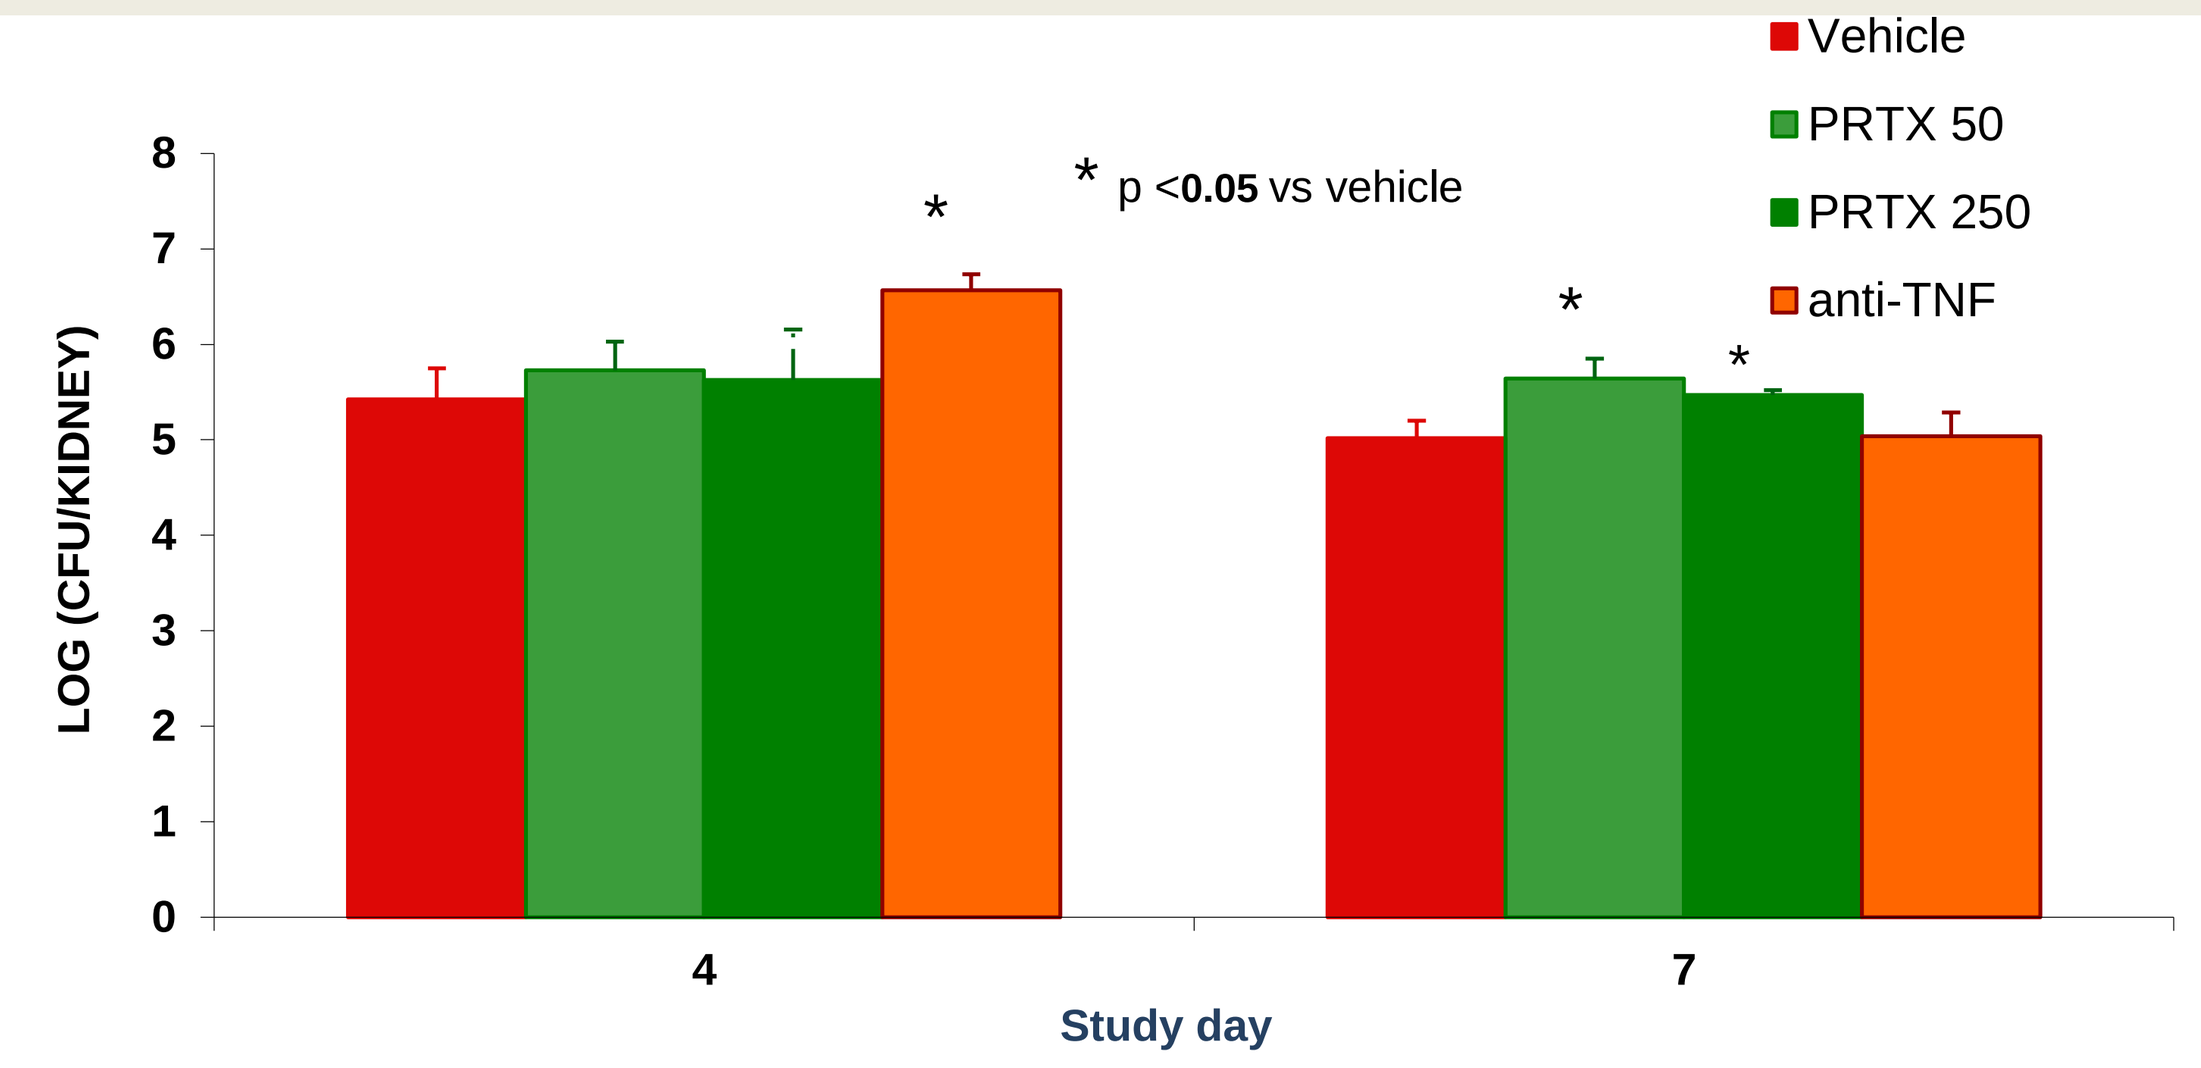
Vehicle
PRTX 50
PRTX 250
anti-TNF
* p <0.05 vs vehicle
LOG (CFU/KIDNEY)
Study day

CONCLUSIONS

1. PRTX-100 (staphylococcal protein A) at doses which equal highest human PRTX-100 exposures in clinical trials (50 µg/kg) or exceed them (250 µg/kg), and which are effective in mouse CIA, do not effect lethality or morbidity in listeria and candida challenge models.
2. Treatment with etanercept, which binds mouse TNF-alpha, or with anti-mouse TNF, increases lethality and severity in both infectious challenge models.
3. In both models, there was a trend towards greater survival and decreased weight loss with 250 µg/kg of PRTX-100, but this did not reach statistical significance in individual experiments, due to their limited group size.
4. These data further differentiate the mechanism of action of PRTX-100 from that of anti-TNF therapeutics.