Presentation #49
ASM-ESCMID 9/5/2019

In Vitro Activity of the Novel Antibacterial ACX-362E against *Clostridioides difficile*

B. Murray,[1] A. Marra,[1] M. H. Silverman,[2] C. Pillar,[1] and D. Shinabarger[1]

[1]Micromyx, Inc. Kalamazoo, MI, [2]Acurx LLC, White Plains, NY

Micromyx, LLC
amarra@micromyx.com

ABSTRACT

Background: ACX-362E is a novel DNA polymerase IIIC inhibitor in clinical development for the treatment of *Clostridioides difficile* infections. Therefore, it is important to show that toxin-producing and epidemic strains of C. difficile are susceptible to ACX-362E. In this study, the in vitro activity of ACX-362E was evaluated against a panel of 104 isolates of C. difficile, including those with relevant ribotypes (e.g. 027) and those producing Toxin A/B.

Methods: The agar dilution method per CLSI (M11-A8) was used to determine MIC values in this study; metronidazole (MET), vancomycin (VAN) and fidaxomicin (FDX) were the comparator agents (M100-S28). Of the 104 C. difficile isolates tested, 30 were of different ribotypes and 36 produced at least one of the A/B toxins.

Results: The activity of ACX-362E and comparators against 104 C. difficile isolates is summarized below:

	ACX-362E	MTZ	VAN	FDX
MIC range:	1 - 8	0.25 - 16	0.5 - 4	0.015 - 1
MIC_{50}:	4	0.5	1	0.12
MIC_{90}:	4	1	2	0.25

The $MIC_{50/90}$ values for ACX-362E against the panel of *C. difficile* isolates overall was 4/4 µg/mL; when parsed against those characterized by ribotype (n=30) or those that were toxin producers (n=36), the $MIC_{50/90}$ values were 2/4 and 4/4 µg/mL, respectively. The comparators metronidazole, vancomycin and fidaxomicin had MIC_{90} values of 1, 2 and 0.25 µg/mL, respectively, against this set of *C. difficile* isolates overall, and similar MIC_{90} values were observed against the 30 different ribotypes or the 36 isolates that produced toxin.

Conclusions: Overall, the results of this study indicated activity for ACX-362E that was similar to that of the comparators evaluated, with a narrow MIC range, even with 30 isolates being of different ribotypes and another 36 isolates being toxin-producers. In addition, four isolates of the epidemic strain ribotypes 027 and 078 demonstrated ACX-362E sensitivities similar to those of other ribotypes. These data support the continued development of ACX-362E, the first DNA polymerase IIIC to enter human trials, for the treatment of *C. difficile* infections.

INTRODUCTION

- The Gram-positive anaerobic spore-forming bacterium *Clostridioides difficile* is the most frequently-reported nosocomial pathogen, recently surpassing *Staphylococcus aureus* for infections in the US (1).
- A 2011 surveillance study reported that 453,000 cases and 29,000 deaths were associated with *C. difficile*; approximately 25% of these were community-acquired (2).
- Relapse/recurrence post-treatment with therapies such as metronidazole (MET) and vancomycin (VAN) are frequent (3).
- The severity and prevalence of these infections have caused the CDC to identify *C. difficile* as an “urgent threat” (4).
- ACX-362E is a dichlorobenzyl purine analog with a unique mechanism of action that binds and inhibits DNA polymerase IIIC from aerobic, low G+C Gram-positive bacteria. ACX-362E is being developed for oral treatment of *C. difficile* infections (CDI).
- ACX-362E has been shown to directly inhibit the DNA pol IIIC from *C. difficile*, with a K_i of 0.325 µM (5).
- DNA pol III is absent in Gram-negative bacteria and thus, ACX-362E lacks activity against most of the organisms that make up the colonic flora, enhancing its profile as a selective treatment for CDI.

OBJECTIVE

- In this study the activity of ACX-362E was evaluated against 104 clinical isolates of *C. difficile,* including toxin-producing and epidemic strains.

METHODS

- The *C. difficile* isolates evaluated in this study consisted of 104 non-duplicate clinical isolates collected between 2003 and 2015 in the US and included 35 toxigenic strains and 31 strains of different ribotypes.
- MIC determinations were conducted by agar dilution using supplemented Brucella agar in accordance with CLSI guideline M11-A8 and CLSI supplement M100-S28.
- Relevant quality control isolates from the ATCC were included during testing. MIC values for QC isolates were within established quality control ranges for all drugs.

RESULTS

- As summarized in **Table 1**, against these isolates overall, ACX-362E demonstrated an MIC range of 1 to 8 µg/mL, with an MIC_{50} of 4 µg/mL and an MIC_{90} of 4 µg/mL.
- By comparison, MET had an MIC range of 0.25 to 16 µg/mL, with an $MIC_{50/90}$ of 0.5/1 µg/mL.
- The comparator VAN showed an MIC range of 0.5 to 4 µg/mL with an $MIC_{50/90}$ of 1/2 µg/mL.
- FDX displayed an MIC_{50} of 0.12 µg/mL, an MIC_{90} of 0.25 µg/mL and an MIC range of 0.015 to 1 µg/mL.
- The activity of ACX-362E against the toxigenic isolates was similar to that of the overall panel, with $MIC_{50/90}$ values of 4/4 µg/mL and a narrow MIC range of 1 to 4 µg/mL.
- MET and VAN had $MIC_{50/90}$ values of 0.5/2, 1/2 µg/mL, respectively, against the toxigenic isolates. FDX was most active with $MIC_{50/90}$ values of 0.12/0.25 µg/mL against these isolates.
- Against 30 isolates of *C. difficile* of different ribotypes, ACX-362E displayed $MIC_{50/90}$ values of 2/4 µg/mL; corresponding values for MET and VAN were 0.25/1 and 1/2 µg/mL, respectively. FDX demonstrated $MIC_{50/90}$ values of 0.06/0.12 µg/mL against this set of isolates.
- The histograms in **Figure 1** show the activity of ACX-362E, MET, VAN and FDX against all 104 *C. difficile* isolates (A.); the 30 isolates of different ribotypes (B.); and the 36 toxin-producing isolates (C.).
- MIC values for ACX-362E, MET, VAN and FDX against the 30 clinical isolates of *C. difficile* of different ribotypes evaluated in this study are shown in **Table 2**.
- Against the epidemic ribotypes 027 and 078, ACX-362E exhibited MIC values of 1 to 2 µg/mL, in line with the activity of this drug against the other isolates in this study.
- MET and VAN had similar MIC values against isolates with the 027 and 078 ribotypes, whereas MIC values for FDX were 0.03 – 0.06 µg/mL.

RESULTS

Table 1. Summary of activity (µg/mL) of ACX-362E, MET, VAN and FDX against 36 toxin-producing isolates, 30 isolates of different ribotypes and other isolates of *C. difficile*

Drug	Type	N	MIC Range	MIC_{50}	MIC_{90}
ACX-362E	Overall	104	1 - 8	4	4
MET			0.25 - 16	0.5	1
VAN			0.5 - 4	1	2
FDX			0.015 - 1	0.12	0.25
ACX-362E	Toxigenic	36	1 - 4	4	4
MET			0.25 - 4	0.5	2
VAN			0.5 - 2	1	2
FDX			0.015 - 0.25	0.12	0.25
ACX-362E	Different ribotypes	30	1 - 8	2	4
MET			0.25 - 16	0.25	1
VAN			0.5 - 2	1	2
FDX			0.015 - 0.12	0.06	0.12

Figure 1. Activity of ACX-362E, MET, VAN and FDX against all 104 *C. difficile* isolates (A.), isolates of different ribotypes (B.), and toxin-producing strains (C.)

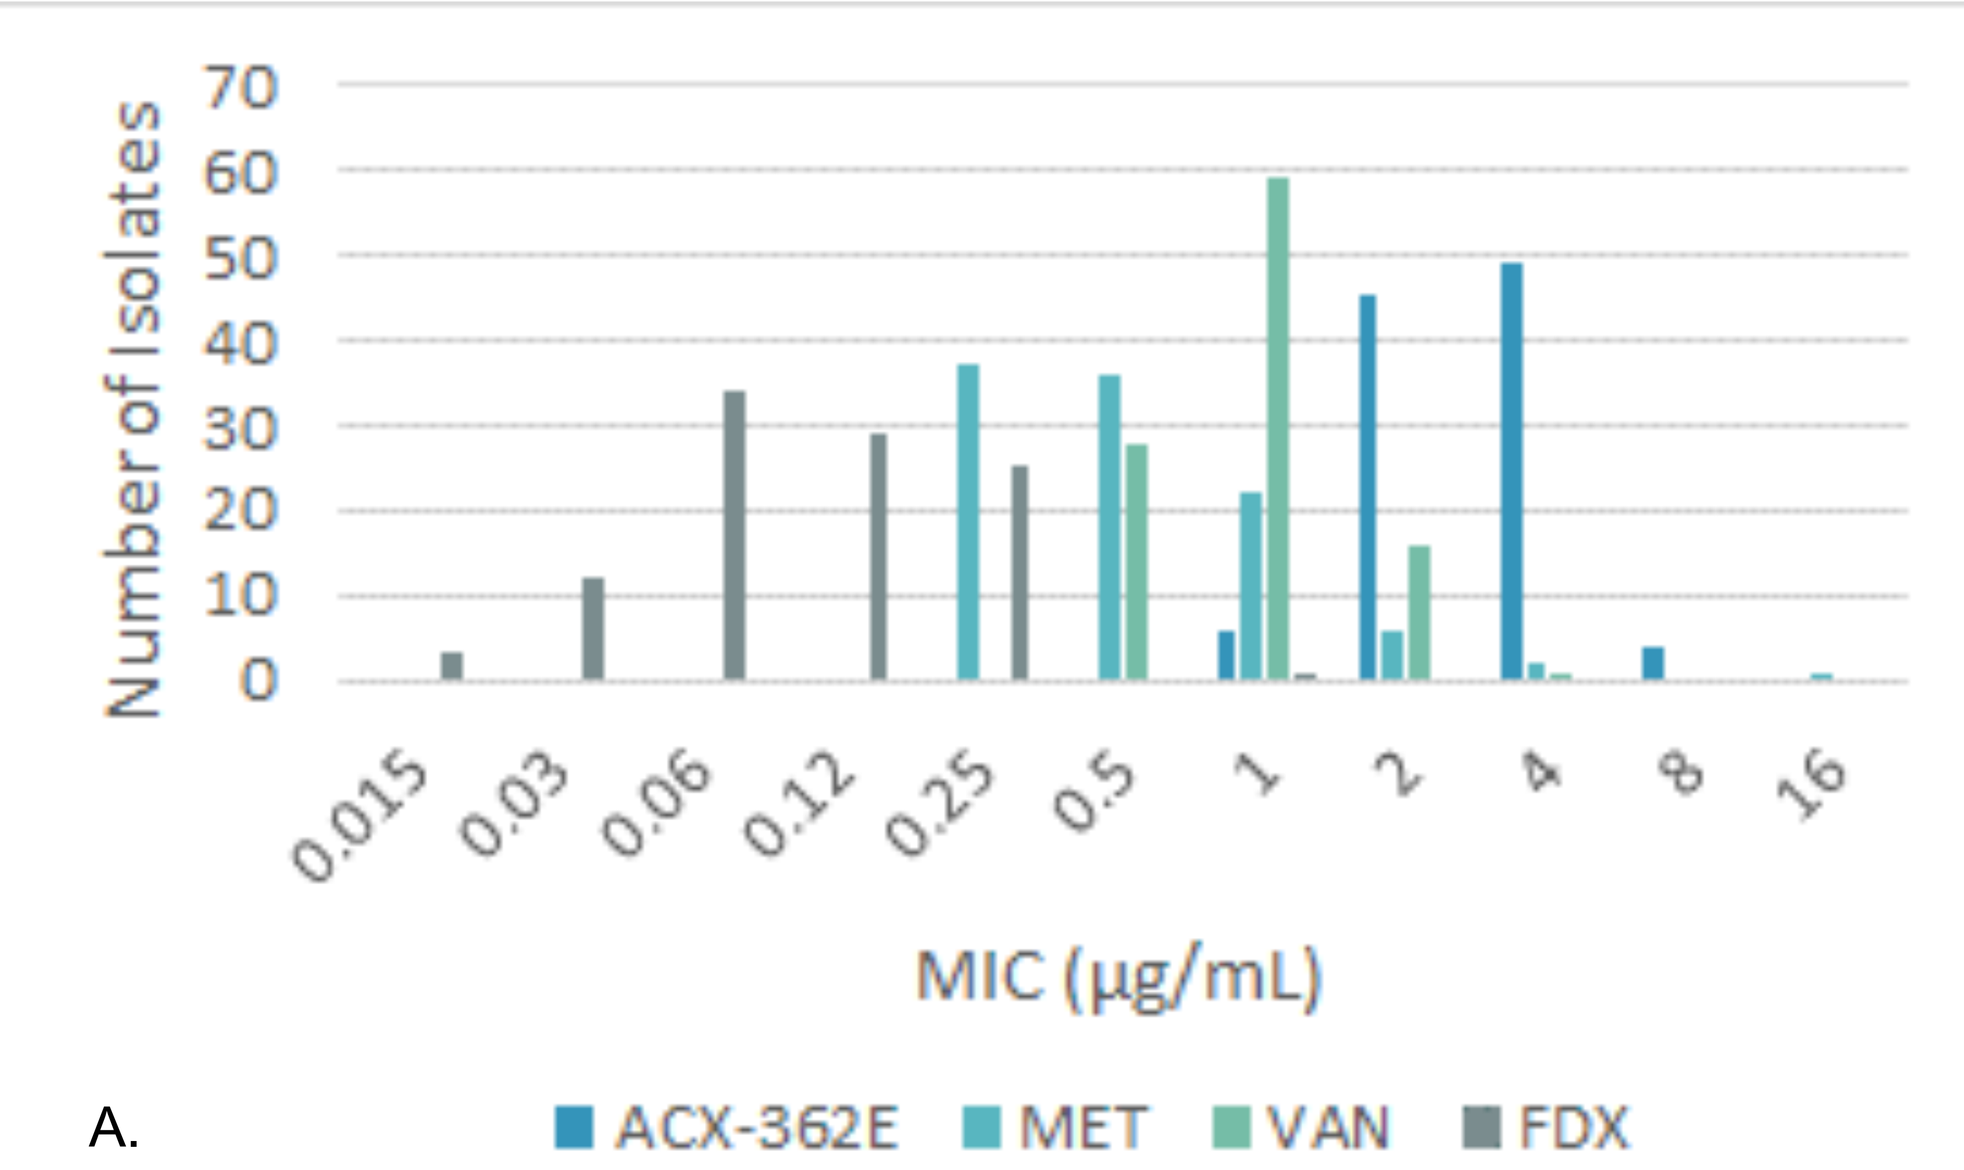

Number of Isolates
70
60
50
40
30
20
10
0
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
MIC (µg/mL)
ACX-362E
MET
VAN
FDX

A.

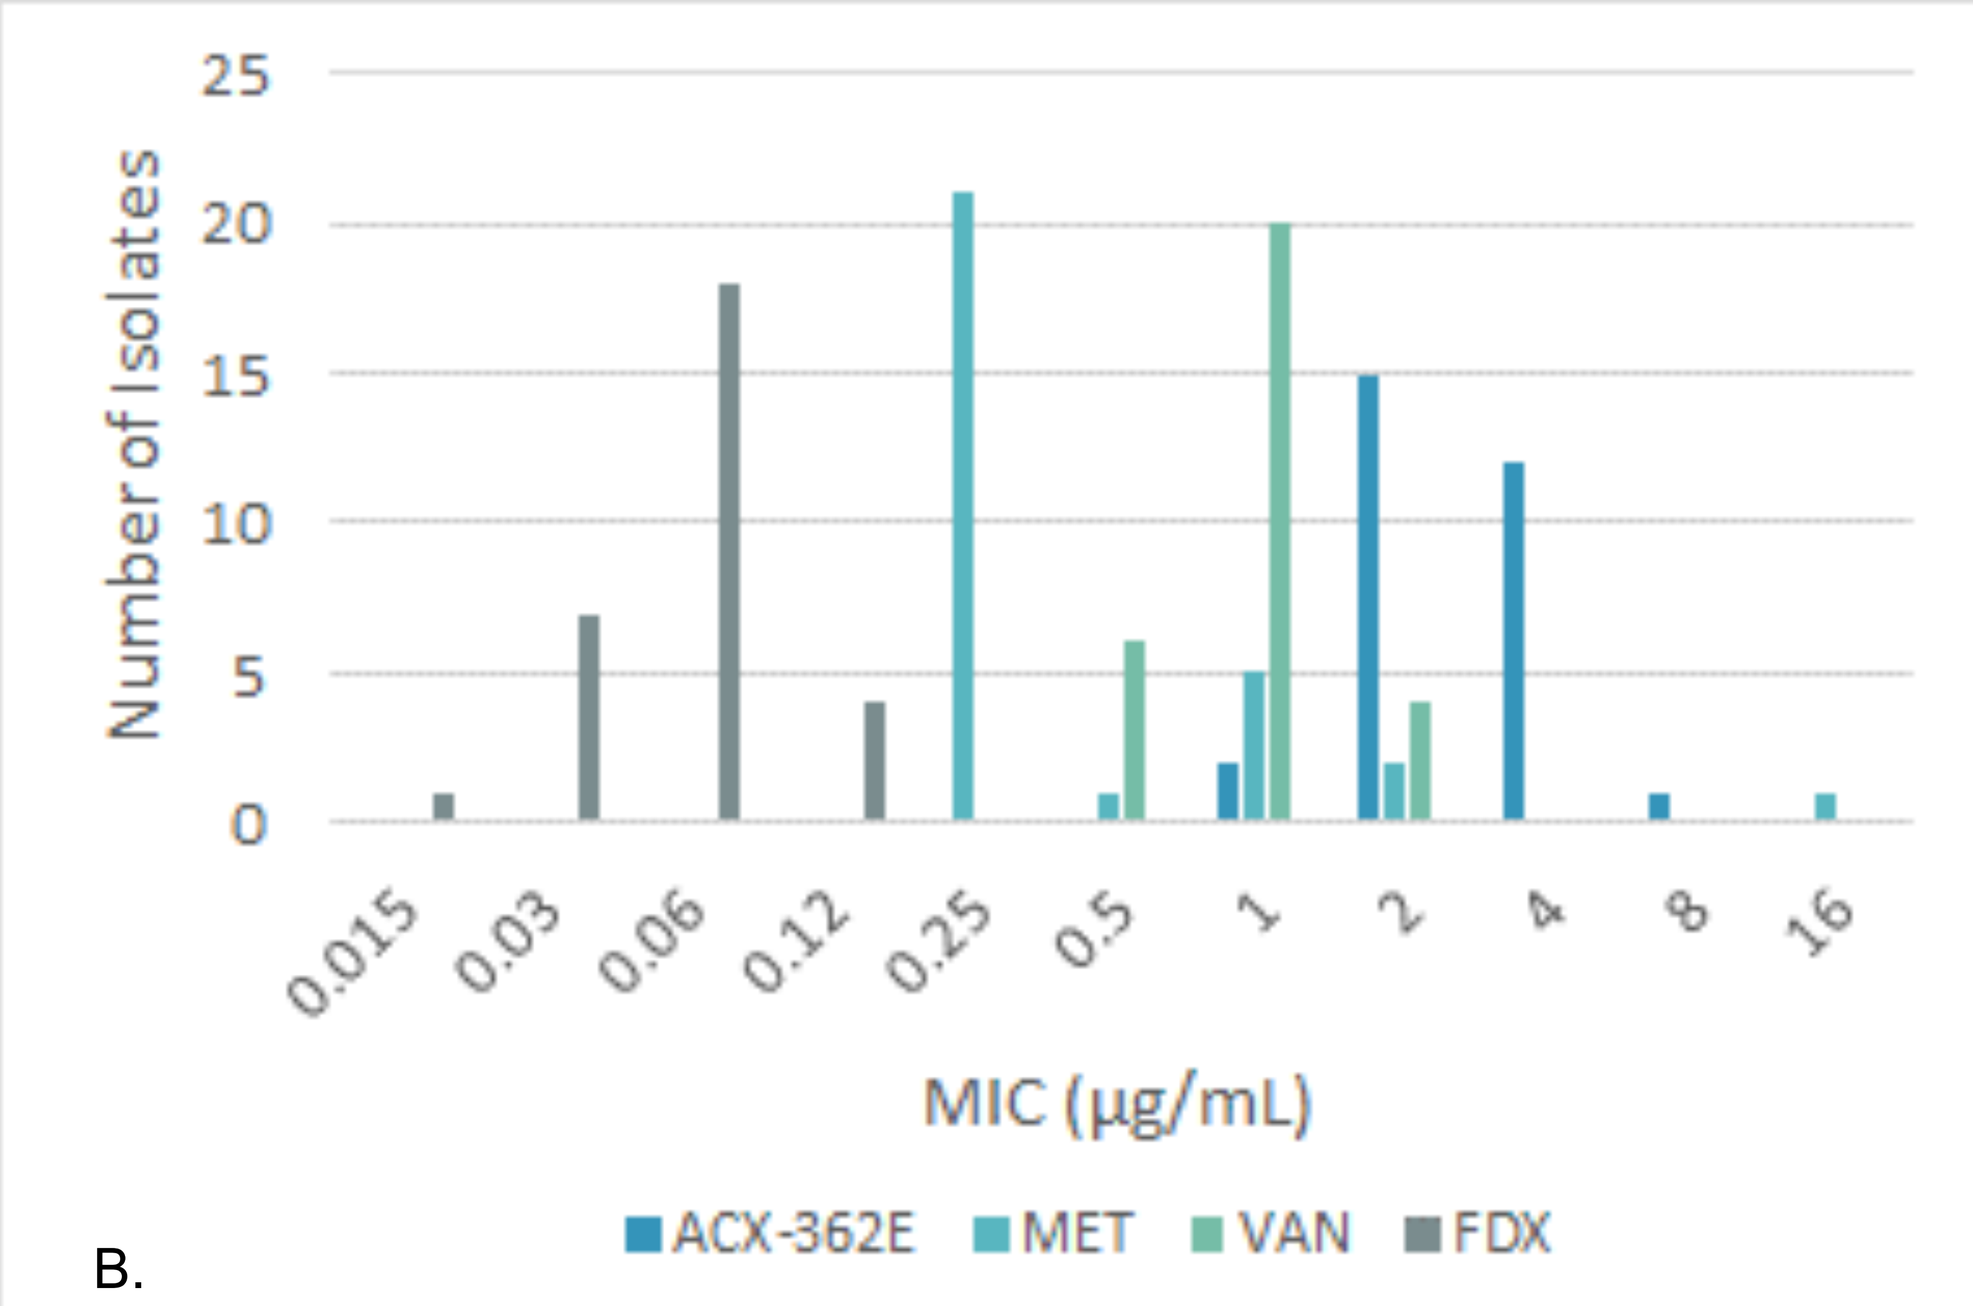

Number of Isolates
25
20
15
10
5
0
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
MIC (µg/mL)
ACX-362E
MET
VAN
FDX

B.

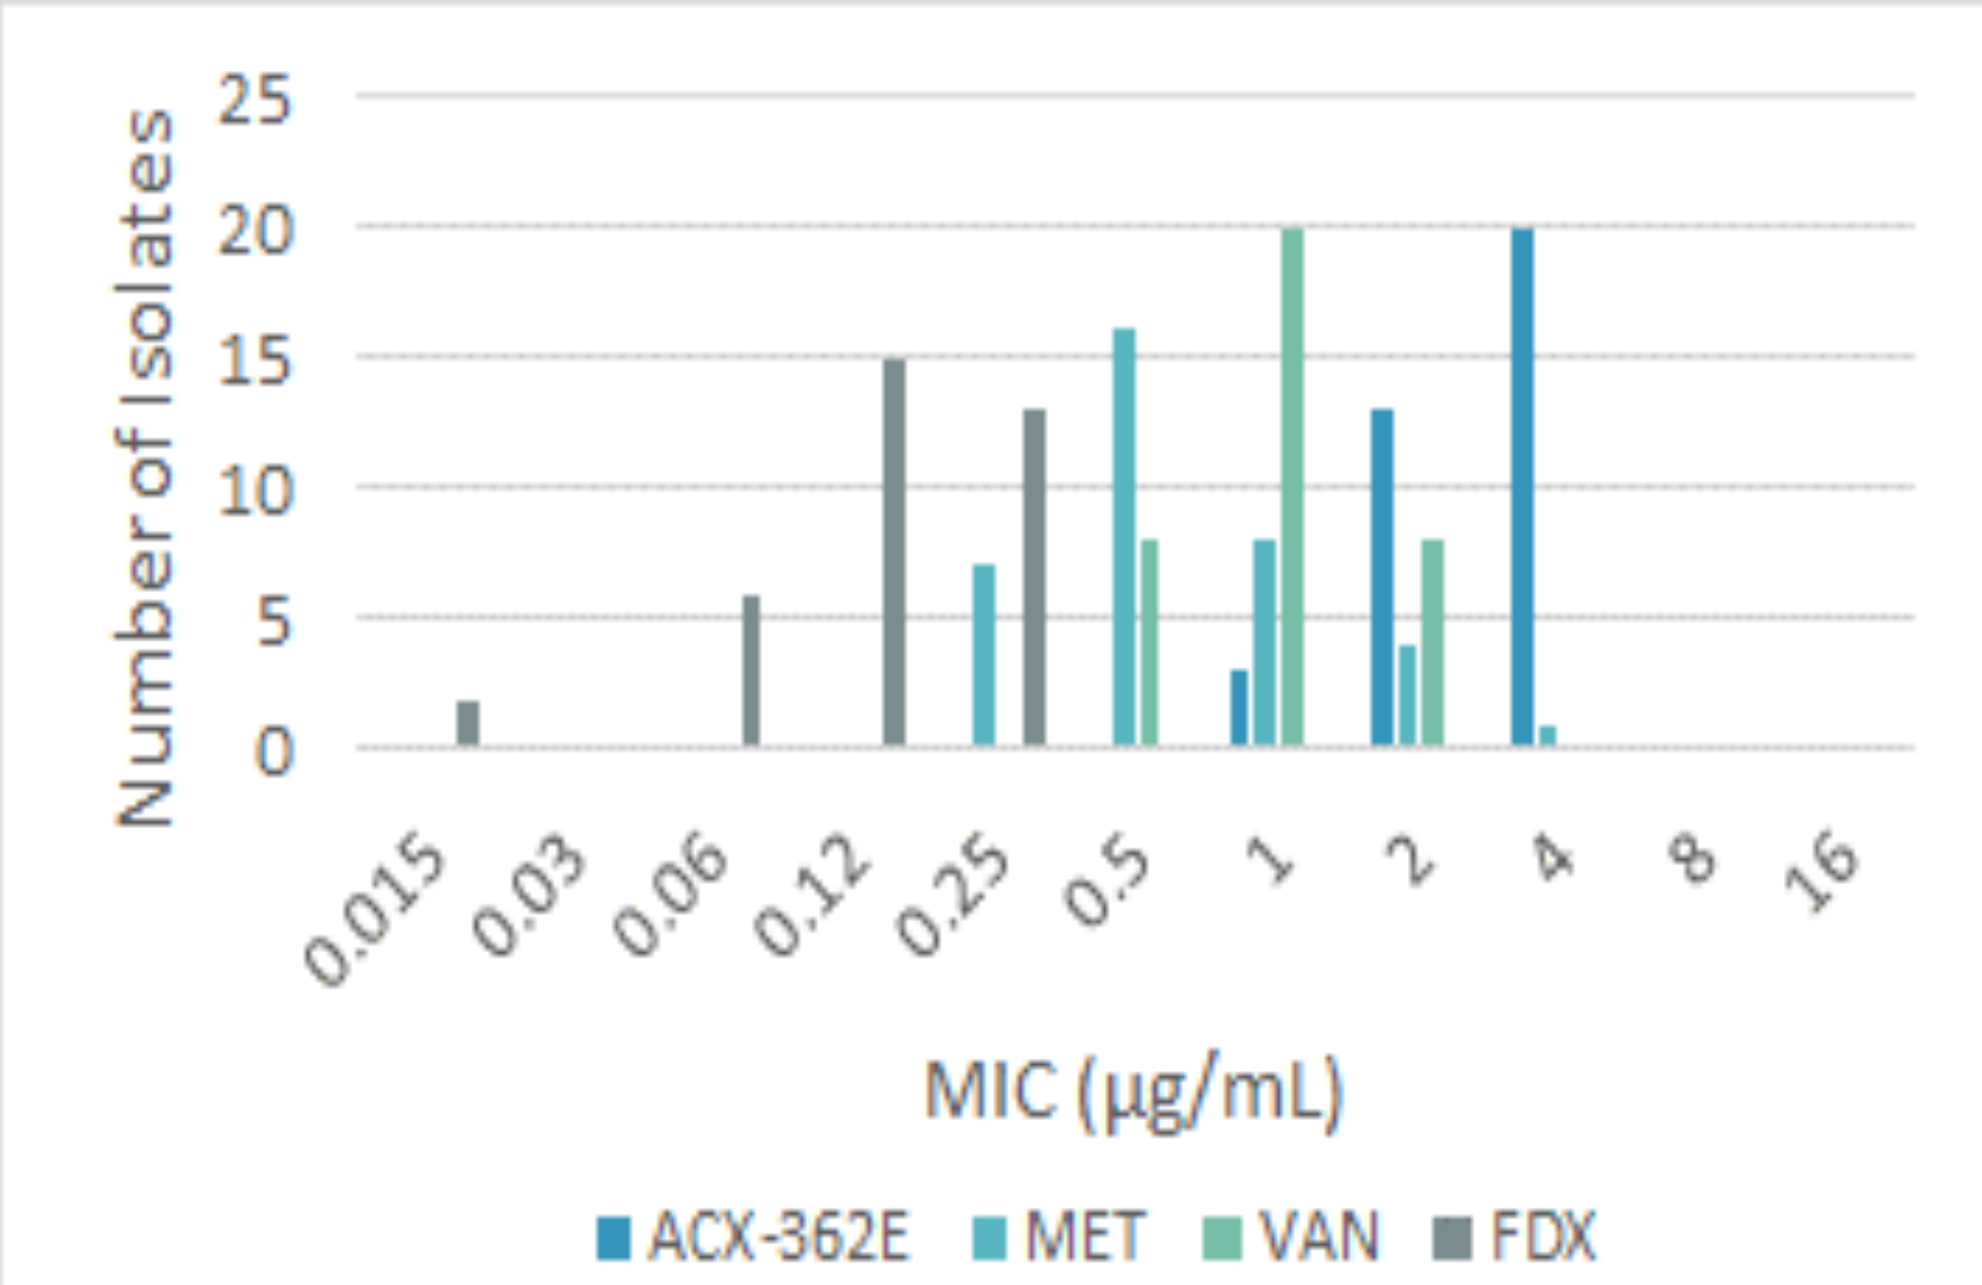

Number of Isolates
25
20
15
10
5
0
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
MIC (µg/mL)
ACX-362E
MET
VAN
FDX

C.

Table 2. Activity of ACX-362E, MET, VAN and FDX against 30 isolates of *C. difficile* of different ribotypes

MMX No.	Type	MIC (µg/mL)			
		ACX-362E	MET	VAN	FDX
8260	ribotype 017	4	0.25	0.5	0.03
8261	ribotype 014	2	0.25	0.5	0.03
8262	ribotype 012	2	0.25	1	0.06
8263	ribotype 015	2	0.25	1	0.06
8264	ribotype 027	2	1	1	0.06
8265	ribotype 027	2	1	0.5	0.06
8266	ribotype 255	4	0.25	0.5	0.06
8267	ribotype 106	4	0.25	0.5	0.12
8268	ribotype 020	4	0.5	1	0.12
8269	ribotype 012	2	0.25	1	0.06
8270	ribotype 027	1	1	2	0.06
8271	ribotype 027	1	2	2	0.06
8272	ribotype 255	2	0.25	1	0.06
8273	ribotype 015	2	0.25	1	0.03
8274	ribotype 076	4	0.25	1	0.06
8275	ribotype 027	2	2	1	0.03
8276	ribotype 023	2	0.25	1	0.06
8277	ribotype 023	4	0.25	1	0.06
8279	ribotype 005	2	0.25	2	0.03
8280	ribotype 023	4	0.25	1	0.06
8281	ribotype 010	8	16	1	0.12
8282	ribotype 017	4	0.25	1	0.03
8283	ribotype 015	2	0.25	1	0.06
8284	ribotype 345	4	0.25	1	0.06
8285	ribotype 010	4	0.25	1	0.06
8286	ribotype 001	4	0.25	1	0.03
8287	ribotype 001	4	1	1	0.015
8288	ribotype 018	2	1	0.5	0.06
8289	ribotype 070	2	0.25	2	0.12
8290	ribotype 078	2	0.25	1	0.06

CONCLUSIONS

- The activity of the novel agent ACX-362E was evaluated in comparison to MET, VAN and FDX against 104 *C. difficile* clinical isolates, of which ~30% were of different ribotypes and another 35% were toxigenic.
- The activity of ACX-362E was similar against the overall set of 104 isolates, the set of toxin-producing isolates, and the set of isolates of different ribotypes.
- In addition, isolates of the epidemic strain ribotypes 027 and 078 demonstrated ACX-362E sensitivities similar to those of other ribotypes.

REFERENCES

1) Evans, CT and Safdar, N. 2015. *Clin Infect Dis*. 60(52): 566 – 71.
2) Leffler, DA and Lamont, JT. 2015. *N Engl J Med*. 372: 1539 – 1548.
3) Jung KS,et al. 2010. *Gut Liver* 2010;4(3): 332-337.
4) Centers for Disease Control and Prevention. Antibiotic threats in the United States, 2013.
5) Torti, A, et al 2011. *Curr Enz Inhib*. 7:147 – 153.