Randomized, Double-blind, Placebo-controlled, Adaptive Design Study to Evaluate DM199 for the Treatment of Acute Ischemic Stroke within a 24-hour Treatment Window (ReMEDy2 Trial)

Scott E. Kasner, MD MSCE, Philip M Bath, FRCP FMedSci, John Volpi, MD on behalf of the ReMEDy2 Trial Investigators.

DiaMedica THERAPEUTICS

INTRODUCTION

Human tissue kallikrein (KLK1) is a serine protease that plays a critical role in regulation of microcirculation, blood pressure, and blood flow, and also mitigates inflammation and oxidative stress. KLK1 is the main bradykinin (BK) producing enzyme which activates the BK2 receptor on endothelial cells, leading to the release of nitric oxide and prostacyclin. The BK2 receptor significantly upregulates in response to ischemia, and the potential effectiveness of DM199 in improving cerebral blood flow in the penumbra is attributed to the increased activation of this receptor. Meta-analyses have indicated that treatment with urinary-derived KLK1 is associated with improved neurological outcomes after stroke. **(Figure 1)**.

DM199 is recombinant form of KLK1 being developed for acute ischemic stroke (AIS). In the prior phase 2 ReMEDy1 study of 91 patients with AIS treated within a 24-hour window, DM199 was safe and well-tolerated. Notably, in the subgroup of 46 patients who did not undergo mechanical thrombectomy, 24% (6/25) treated with DM199 achieved an excellent outcome (mRS 0–1) at day 90, in contrast to 9.5% (2/21) in the placebo group, an absolute improvement of 14.5%, suggesting proof of principle for treatment in this window. Additionally, DM199 was associated with fewer recurrent/progressive ischemic strokes: 0% (0/46) treated with DM199 compared to 13.3% (6/45) in the placebo group, an absolute improvement of 13.3% (p=0.012). Taken together, these findings suggest that DM199 not only promotes recovery but may also ameliorate the progression of stroke.

OBJECTIVE

ReMEDy2 (NCT05065216) is a pivotal study of DM199 in AIS patients, focusing on stroke recovery within a 24-hour treatment window for those for whom thrombolysis (tPA/TNK) and/or a catheter-based procedure like MT are not medically appropriate.

Figure 1. Rationale for DM199 Augmentation Therapy in AIS

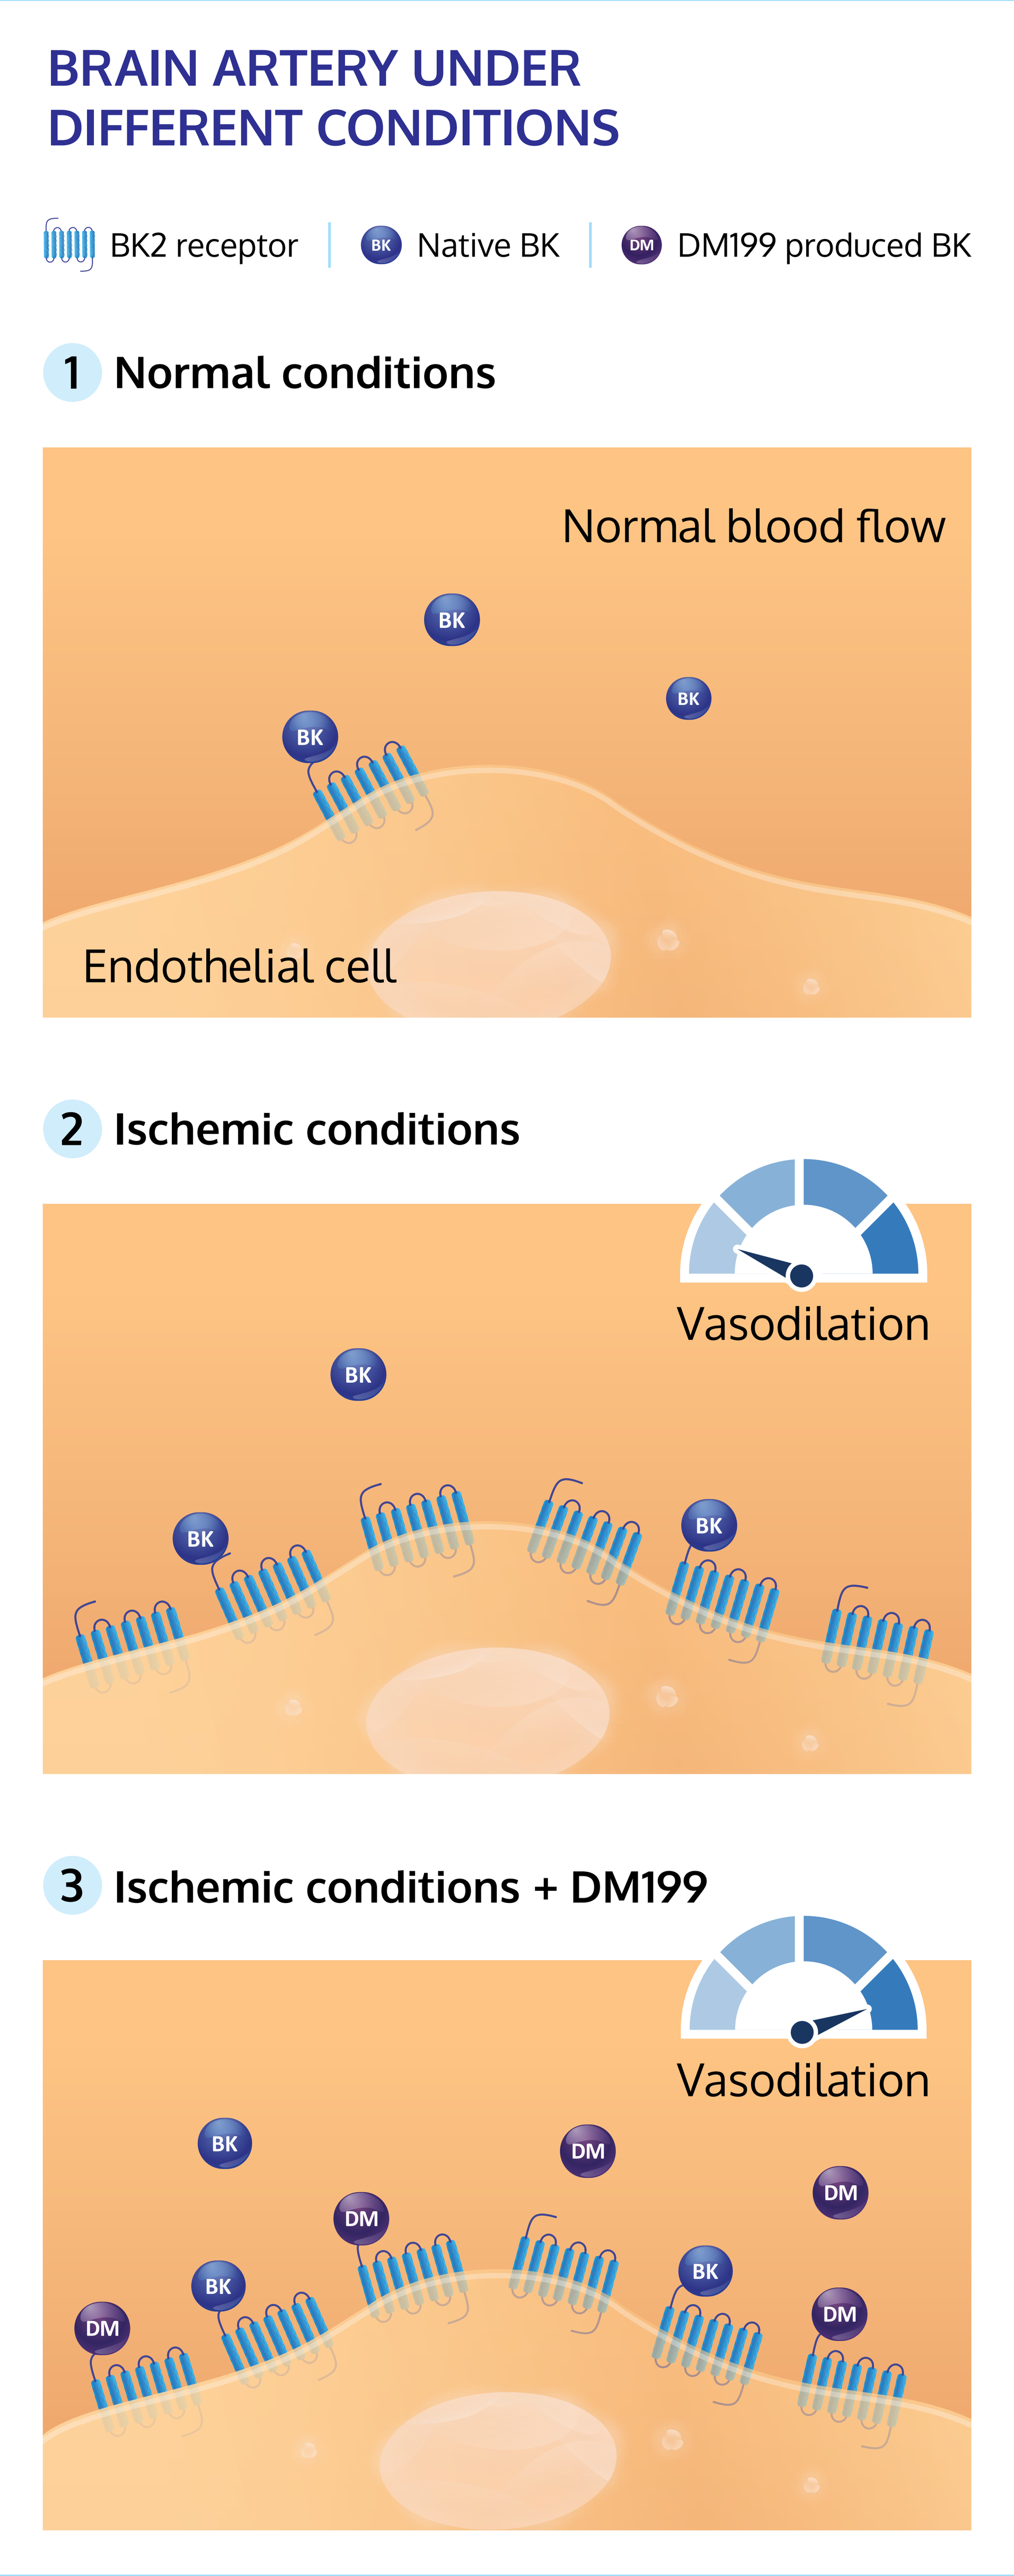
BRAIN ARTERY UNDER DIFFERENT CONDITIONS
BK2 receptor
BK Native BK
DM DM199 produced BK
1 Normal conditions
Normal blood flow
BK
Endothelial cell
2 Ischemic conditions
Vasodilation
BK
3 Ischemic conditions + DM199
Vasodilation
BK
DM

STUDY DESIGN

- Randomized, double-blind, placebo-controlled phase 2/3 seamless adaptive, multicenter study to evaluate the efficacy, safety and tolerability of DM199 with a 24-hour treatment window **(Figure 2)**.
- Randomized 1:1 to DM199 or placebo, administered by a single intravenous (IV) dose followed by a subcutaneous (SC) dose within 12 hours and then SC doses 2 times per week until Day 22.
- Total duration 90 days.

Figure 2. Study Design

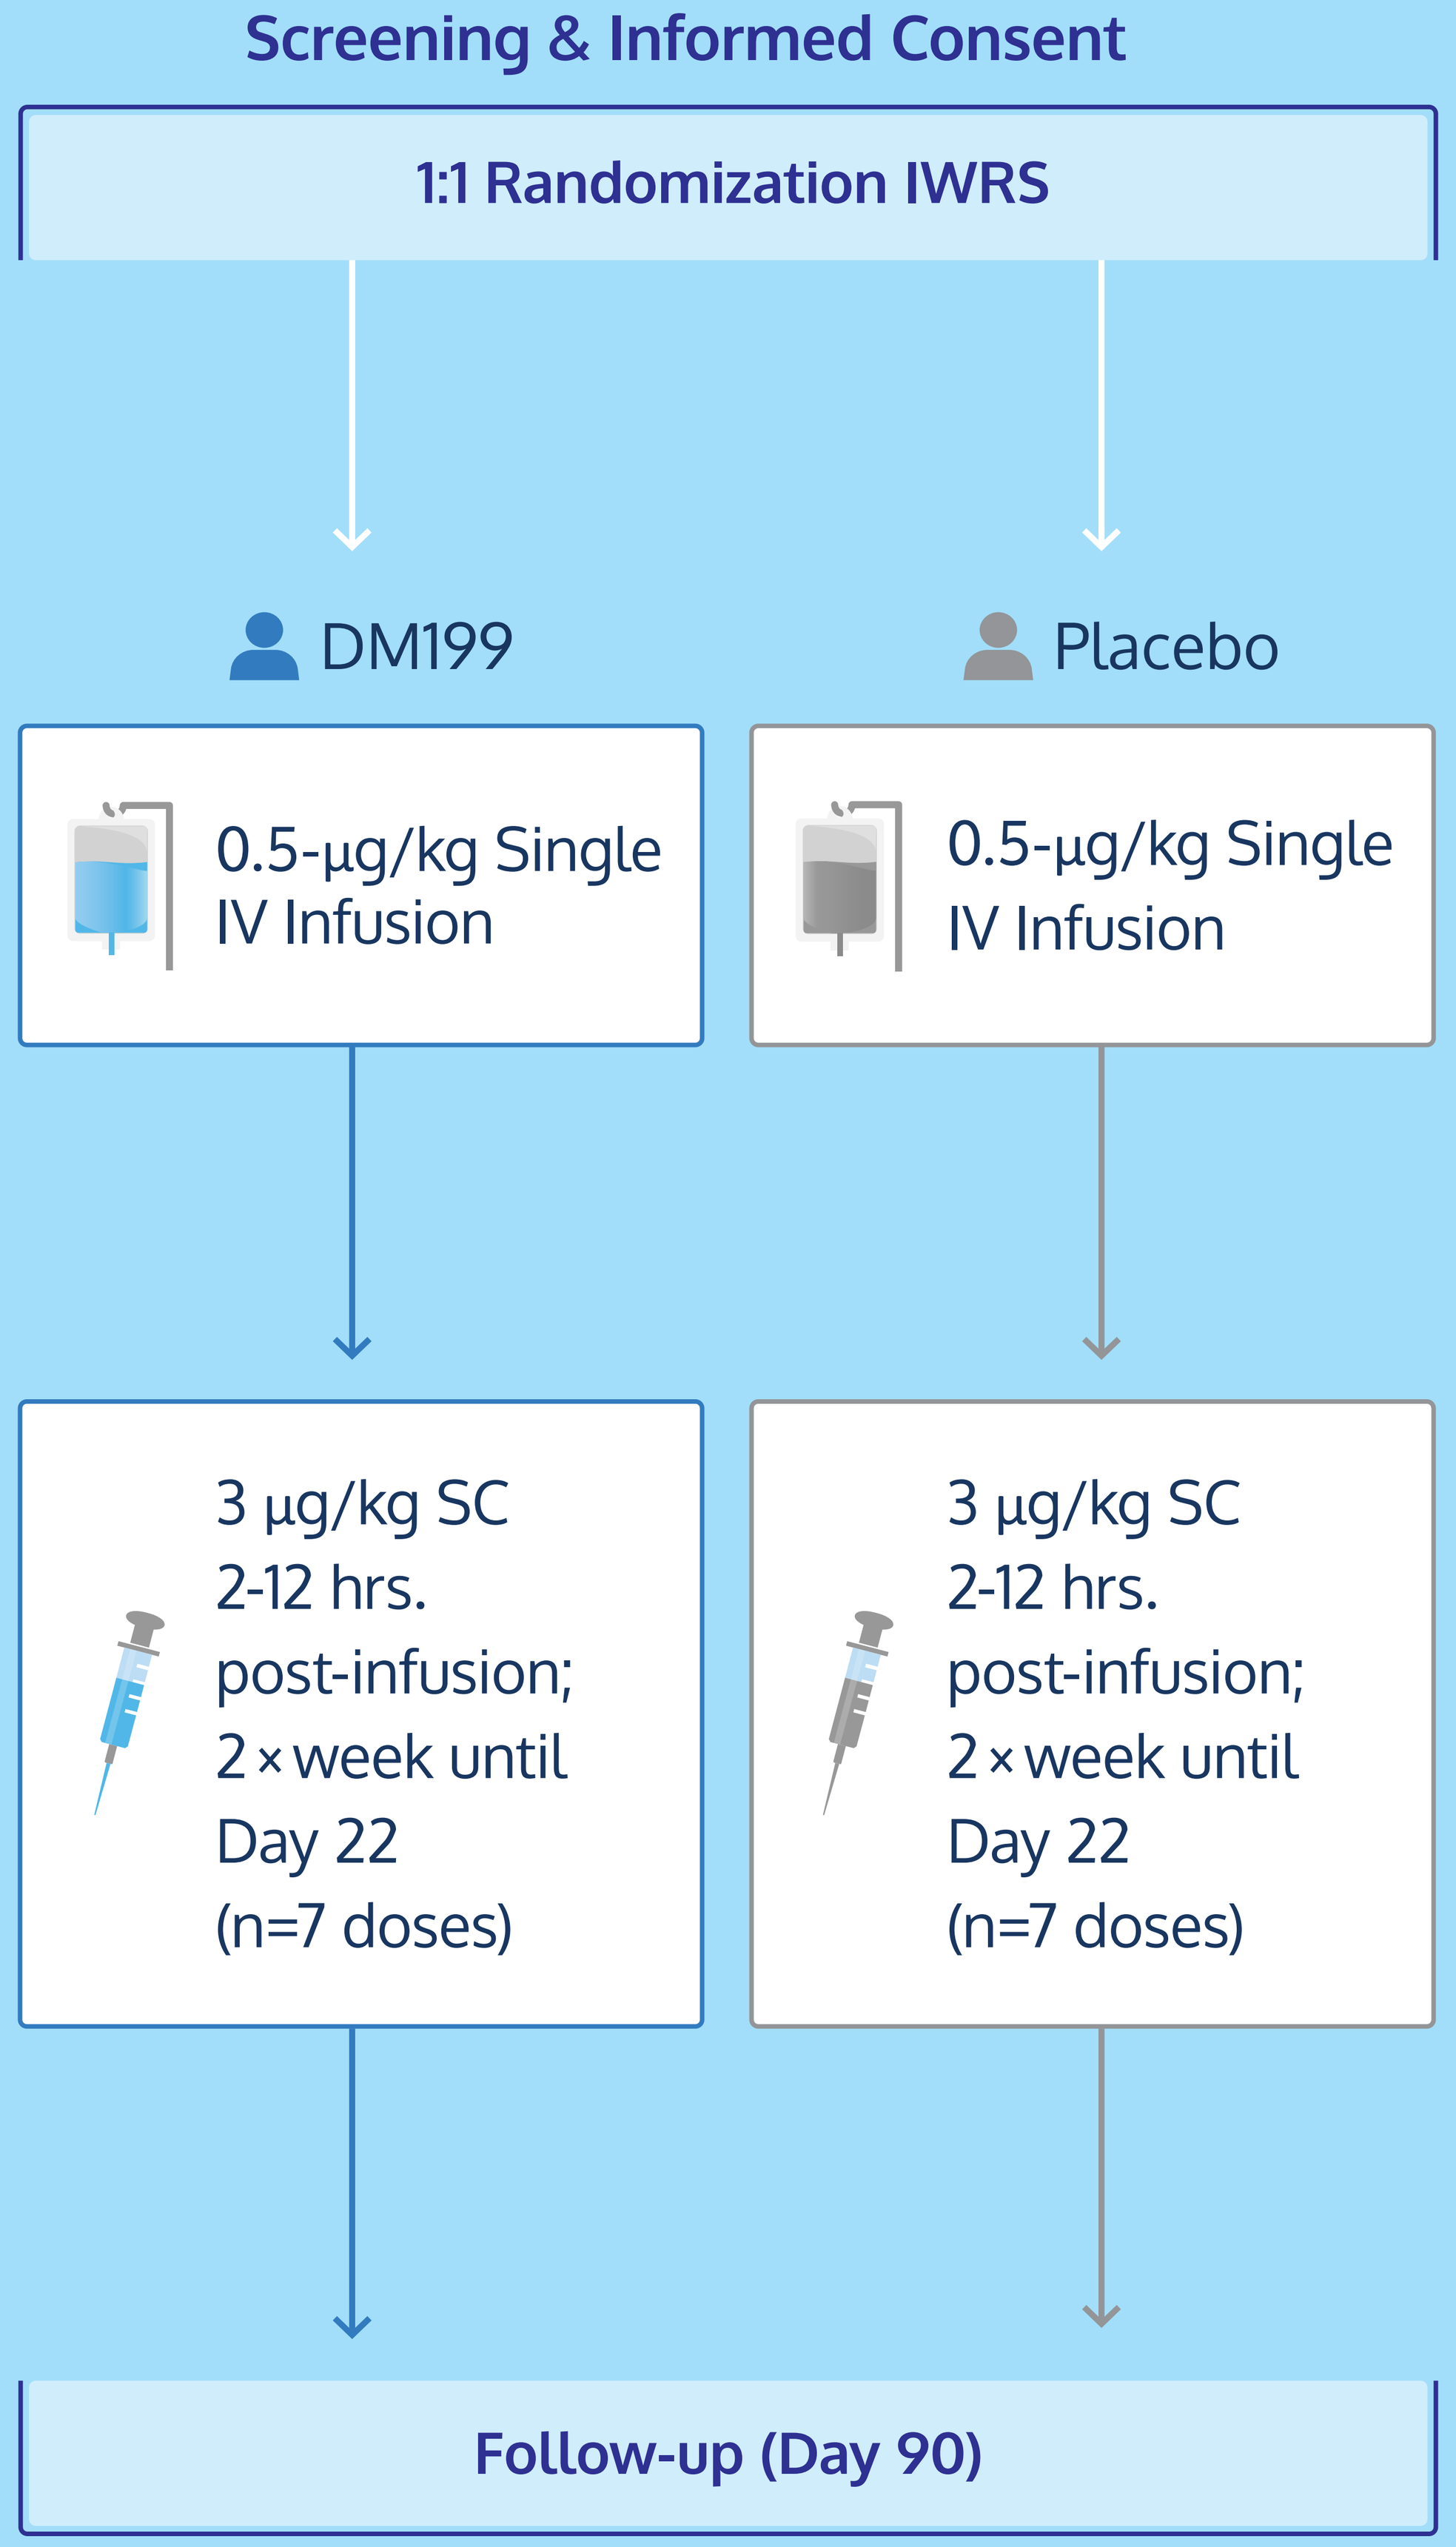
Screening & Informed Consent
1:1 Randomization IWRS
DM199
Placebo
0.5-µg/kg Single IV Infusion
0.5-µg/kg Single IV Infusion
3 µg/kg SC 2-12 hrs. post-infusion; 2 × week until Day 22 (n=7 doses)
3 µg/kg SC 2-12 hrs. post-infusion; 2 × week until Day 22 (n=7 doses)
Follow-up (Day 90)

STUDY POPULATION

- Major selection criteria were based on learnings from the ReMEDy1 study **(Table 1)**.

Table 1. Major inclusion and exclusion criteria

INCLUSION CRITERIA	EXCLUSION CRITERIA
Male or female ≥18 years of age	Received tPA for current AIS
Diagnosed with AIS within 24 hours	Imaging findings consistent with LVO
Not a candidate for tPA	Received or scheduled to receive MT within 24 hours of stroke onset
Not a candidate for MT	ASPECTS score of 0–4
NIHSS ≥5 and ≤15	Unable to complete 24 hour washout of ACE inhibitor prior to DM199 administration
Pre-morbid mRS score of 0-1 prior to stroke	

STUDY ENDPOINTS

- Study outcomes were selected based on results from the ReMEDy1 study with DM199 **(Table 2)**.

Table 2. Outcome measures for ReMEDy2

PRIMARY OUTCOME MEASURE	DEFINITIONS
Efficacy - AIS Recovery	Excellent functional outcomes at Day 90 assessed as mRS score of 0 or 1
Safety	Incidence of adverse events; injection site reactions

SECONDARY OUTCOME MEASURES	
Disability across full spectrum of AIS	Distribution of mRS (shift) scores at Day 90
Participants achieving independent function	mRS score of 0-2 at Day 90
Mortality rate	Mortality event rate (%) over 90 days
Excellent neurological outcome	NIHSS score of 0 or 1 at Day 90
Excellent function independence	Barthel Index score >95 at Day 90
Recurrent AIS	Proportion of patients who experience a recurrent AIS by Day 90 assessed by a new, persistent neurological deficit attributable to cerebrovascular ischemia

STUDY ANALYSIS

- Planned enrollment of approximately 364 patients.
- 90% power at 14% improvement in excellent outcomes.
- Interim analysis planned after 40% of anticipated enrollment have completed 90 day follow-up.

Interim Analysis

- An interim analysis will be conducted after approximately 144 patients complete their Day 90 assessments.
- The study is initially planned to enroll an additional 220 patients in Phase 3 (N=364 total). If sample size re-estimation is deemed appropriate from the interim analysis, a final sample size ranging from 240 to 728 patients is possible.

CONCLUSIONS

- A significant unmet medical need exists for the treatment of AIS patients.
- The need is most particularly in those patients who are not eligible for thrombolysis and acute revascularization treatment.
- Improved flow via enhanced collateral circulation is positively associated with increased salvage of penumbral tissue and negatively associated with the growth of cerebral infarcts.
- In the previous ReMEDy1 Trial, DM199 demonstrated significant improvement in this population, and results from ReMEDy2 are anticipated to confirm these findings.

Presented at the International Stroke Conference (ISC), February 7–9, 2024, Phoenix, AZ + Virtual

CTP16