The glucagon receptor antagonist LGD-6972 reduces fasting and postprandial glucose in a multiple-dose clinical trial

Eric G. Vajda[1], Douglas Logan[2], Kenneth Lasseter[3], Danielle Armas[4], Diane J. Plotkin[5], J.D. Pipkin[1], Yong-Xi Li[2], Rong Zhou[2], David J. Klein[2], Xiaoxiong Wei[2], Stacy Dilzer[3], Lin Zhi[1], and Keith B. Marschke[1]
[1]Ligand Pharmaceuticals Incorporated, La Jolla, CA, [2]Medpace, Inc., Cincinnati, OH, [3]Clinical Pharmacology of Miami, Miami, FL, [4]Celerion, Phoenix, AZ, [5]Diane Plotkin Clinical Development Consultation Services,San Diego, CA

INTRODUCTION

- Glucagon and insulin play counter-regulatory roles in glucose homeostasis.
- In normogylcemic subjects, glucagon is elevated during periods of fasting to mobilize glucose stores. In type 2 diabetes mellitus, glucagon can be inappropriately elevated potentially playing a role in hyperglycemia.
- Antagonism of glucagon action has been proposed as a mechanism to reduce blood glucose levels in type 2 diabetes mellitus.
- We have discovered a novel, selective, orally bioavailable glucagon receptor antagonist and investigated its pharmacokinetics, safety, and pharmacodynamics in a phase 1 multiple dose clinical trial.

CLINICAL TRIAL DESIGN

Primary Objective:
To assess the safety and tolerability of multiple oral doses of LGD-6972 in normal healthy volunteers (NHV) and T2DM subjects.

Secondary Objectives:
- To characterize the steady state pharmacokinetics of LGD-6972.
- To assess the effects of LGD-6972 on glycemic response.

Study Design: This was a multi-center, randomized, double-blind, placebo-controlled multiple ascending dose study in both NHV (n=12) and T2DM subjects (n=36). Subjects were confined to the clinical pharmacology unit and received standardized meals throughout the 14 day dosing period. The NHV cohort was dosed prior to the T2DM cohorts. Dose escalation occurred in the T2DM subjects after review of safety, tolerability, and preliminary PK data from previous dose levels (n=12/cohort; 3-placebo & 9-LGD-6972). T2DM subjects were on a stable dose of metformin and remained on metformin throughout the trial. All subjects were administered once daily placebo or LGD-6972 as a solution formulated with CAPTISOL® prior to morning breakfast.

Pharmacokinetics: Plasma concentrations of LGD-6972 were measured by a validated LC-MS/MS method. A time-exposure profile was measured throughout a 24 hour period on day 1 and day 14 . Additional trough concentrations were measured at several time points to investigate steady state pharmacokinetics and clearance rates.

Pharmacodynamics: Plasma glucose, glucagon, glucagon-like peptide-1, and insulin were measured. In the 10 mg T2DM cohort, an oral glucose tolerance test was performed on day -1 and day 14. 7-point plasma glucose measurements were performed on day -1 and day 14 for all T2DM cohorts.

DEMOGRAPHICS

Normal Healthy Volunteers		LGD-6972 Dose Level	
		Placebo	15 mg
Number of subjects		3	9
Age (yrs)		47.3 (8.7)	42.3 (10.8)
Gender (n)	Female	0	0
	Male	3	9
Ethnicity (n)	Hispanic or Latino	1	3
	Not Hispanic or Latino	2	6
Race (n)	White	1	4
	African American	2	5
BMI (kg/m^2)		27.4 (1.7)	24.4 (2.9)

Type 2 Diabetes Mellitus Subjects		LGD-6972 Dose Level			
		Placebo	5 mg	10 mg	15 mg
Number of subjects		9	9	9	9
Age (yrs)		52.2 (6.2)	55.1 (6.0)	58.9 (6.7)	53.6 (8.7)
Gender (n)	Female	6	2	4	5
	Male	3	7	5	4
Ethnicity (n)	Hispanic or Latino	7	5	6	6
	Not Hispanic or Latino	2	4	3	3
Race (n)	White	8	8	7	6
	African American	1	1	2	3
BMI (kg/m^2)		31.0 (4.7)	33.6 (4.2)	32.0 (4.9)	31.3 (5.1)
Baseline HbA1c (%)		8.5 (0.9)	8.2 (0.7)	8.2 (1.1)	8.5 (0.9)

SAFETY AND PK

There were no discontinuations or serious adverse events during the study. LGD-6972 was well tolerated with no clinically significant or dose dependent changes in hematology, clinical chemistry, urinalysis, ECG, or vital signs. Adverse events were generally of mild or moderate severity (grade 1 or 2). There were no reported cases of hypoglycemia.
Consistent with prior studies, LGD-6972 was well absorbed after oral administration with a long half life ($t_{1/2}$ approx. 50 hours) and steady state pharmacokinetics was achieved 7-10 days after dosing started (data not shown). The PK profile supports once daily dosing.

GLYCEMIC EFFECTS

Figure 1. Fasting plasma glucose (FPG) decreased after administration of 15 mg LGD-6972 in normal healthy volunteers. FPG remained low throughout the 14 day dosing period.

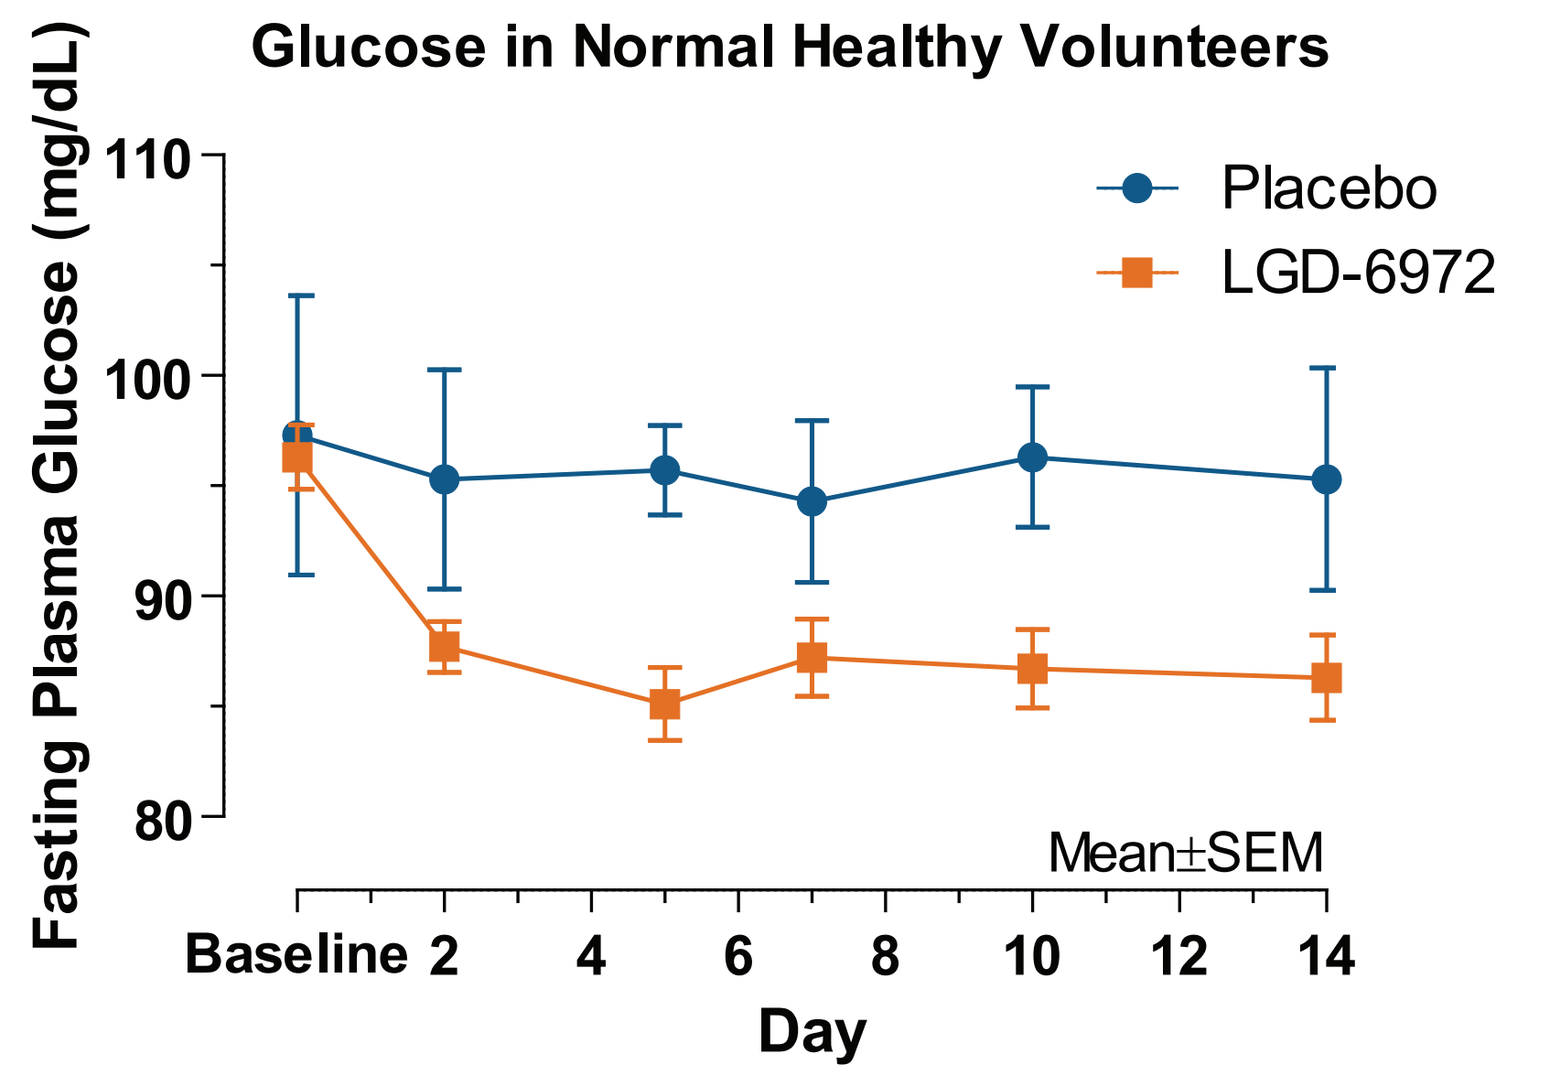
Glucose in Normal Healthy Volunteers
Fasting Plasma Glucose (mg/dL)
Placebo
LGD-6972
Mean±SEM
Day

Figure 2. FPG was reduced in a dose dependent manner in T2DM subjects administered LGD-6972. A maximal reduction of 55 mg/dL was observed in the 15 mg dose. FPG returned to baseline levels after dosing stopped on day 14.

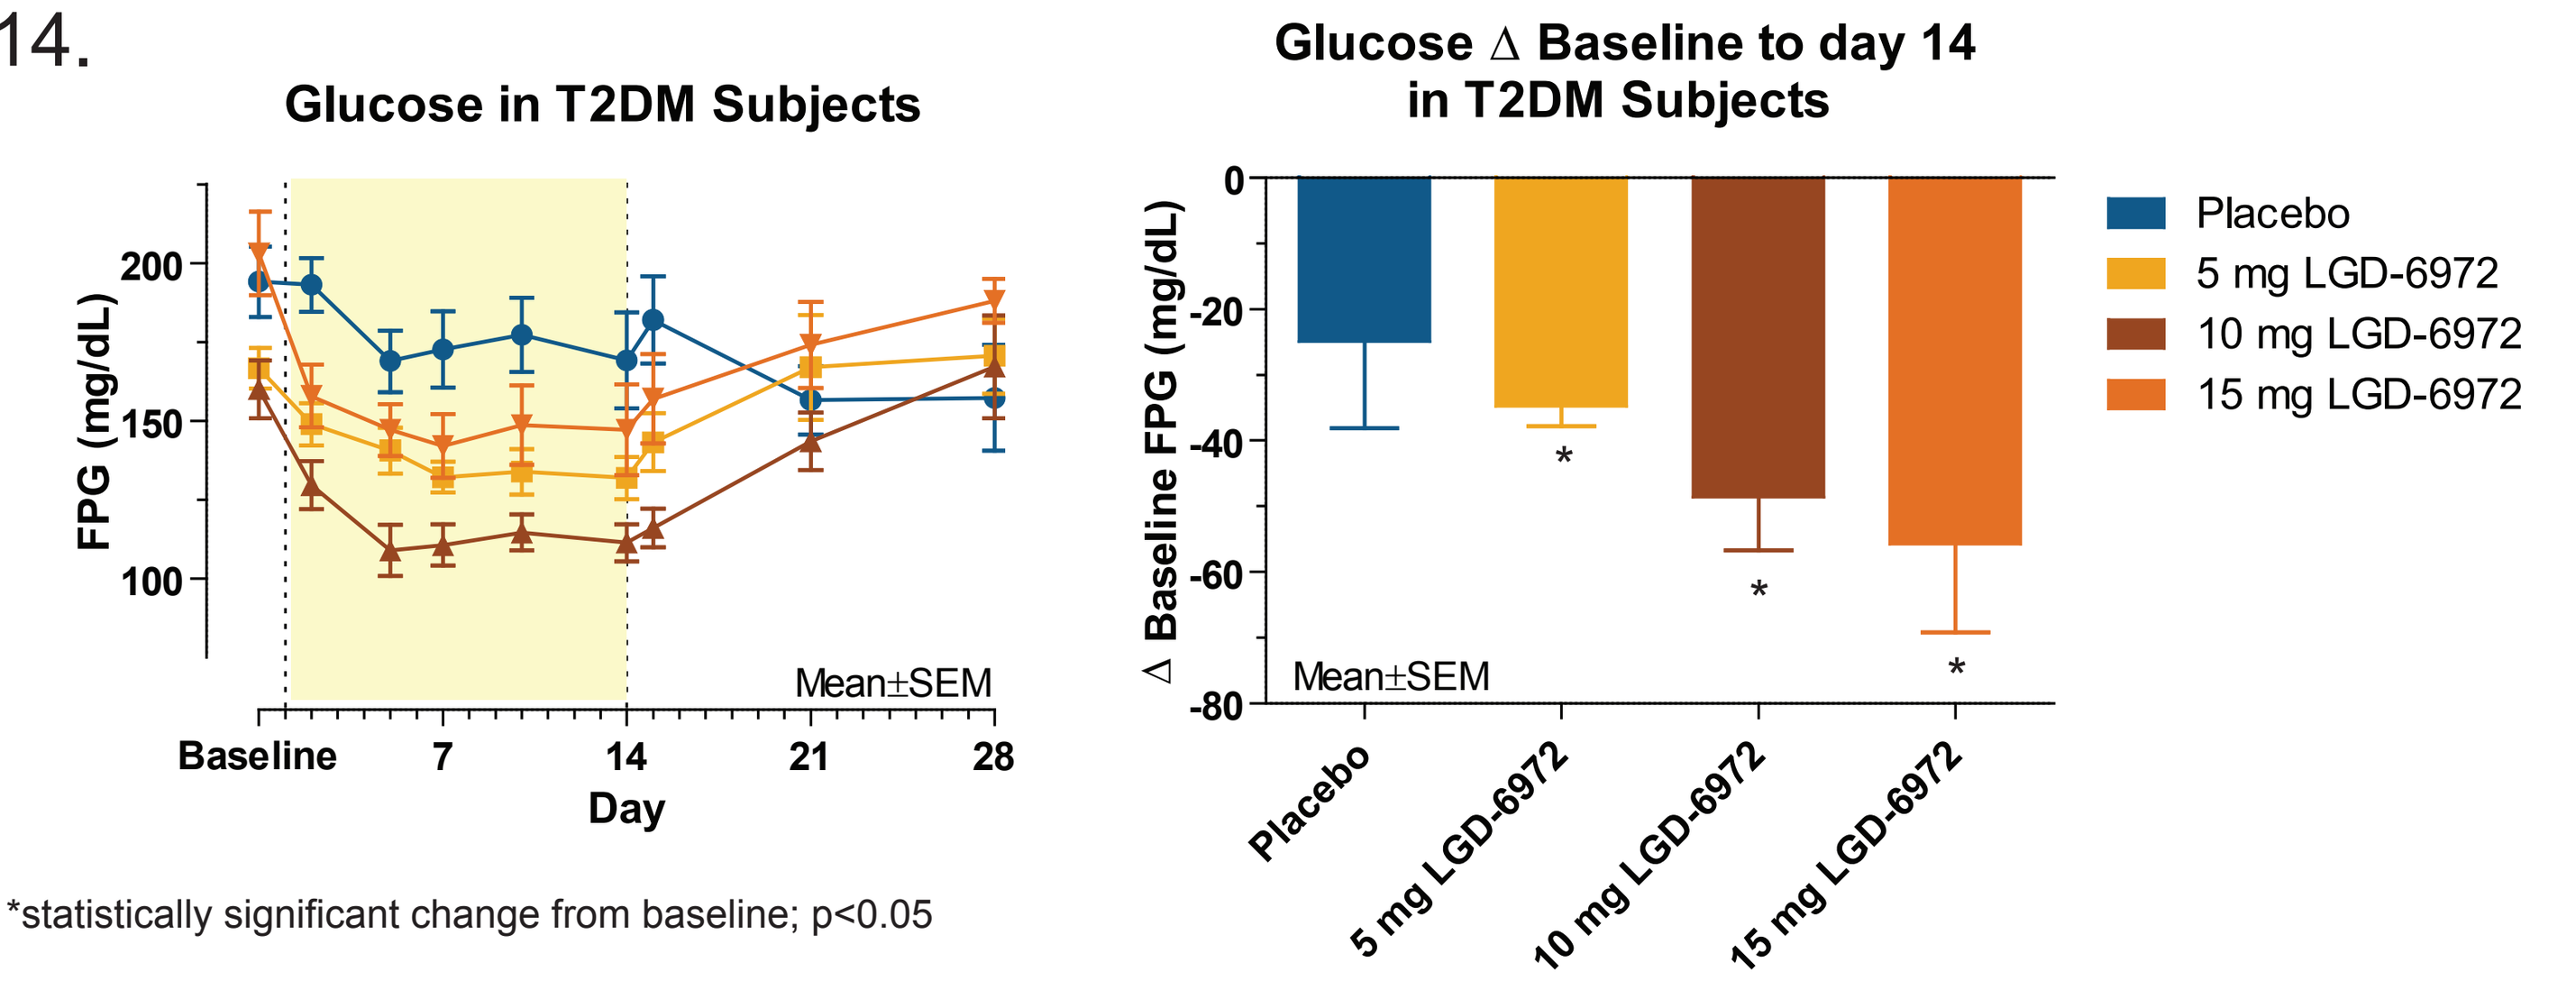
Glucose in T2DM Subjects
FPG (mg/dL)
Mean±SEM
Day
Glucose Δ Baseline to day 14 in T2DM Subjects
Δ Baseline FPG (mg/dL)
Placebo
5 mg LGD-6972
10 mg LGD-6972
15 mg LGD-6972

*statistically significant change from baseline; p<0.05

Figure 3. Seven-point glucose measurements were performed at baseline and on day 14. LGD-6972 reduced glucose in both fasting and postprandial states with a significant change from baseline weighted average glucose (-52.9 mg/dL, p<0.0001) observed in the 15 mg cohort.

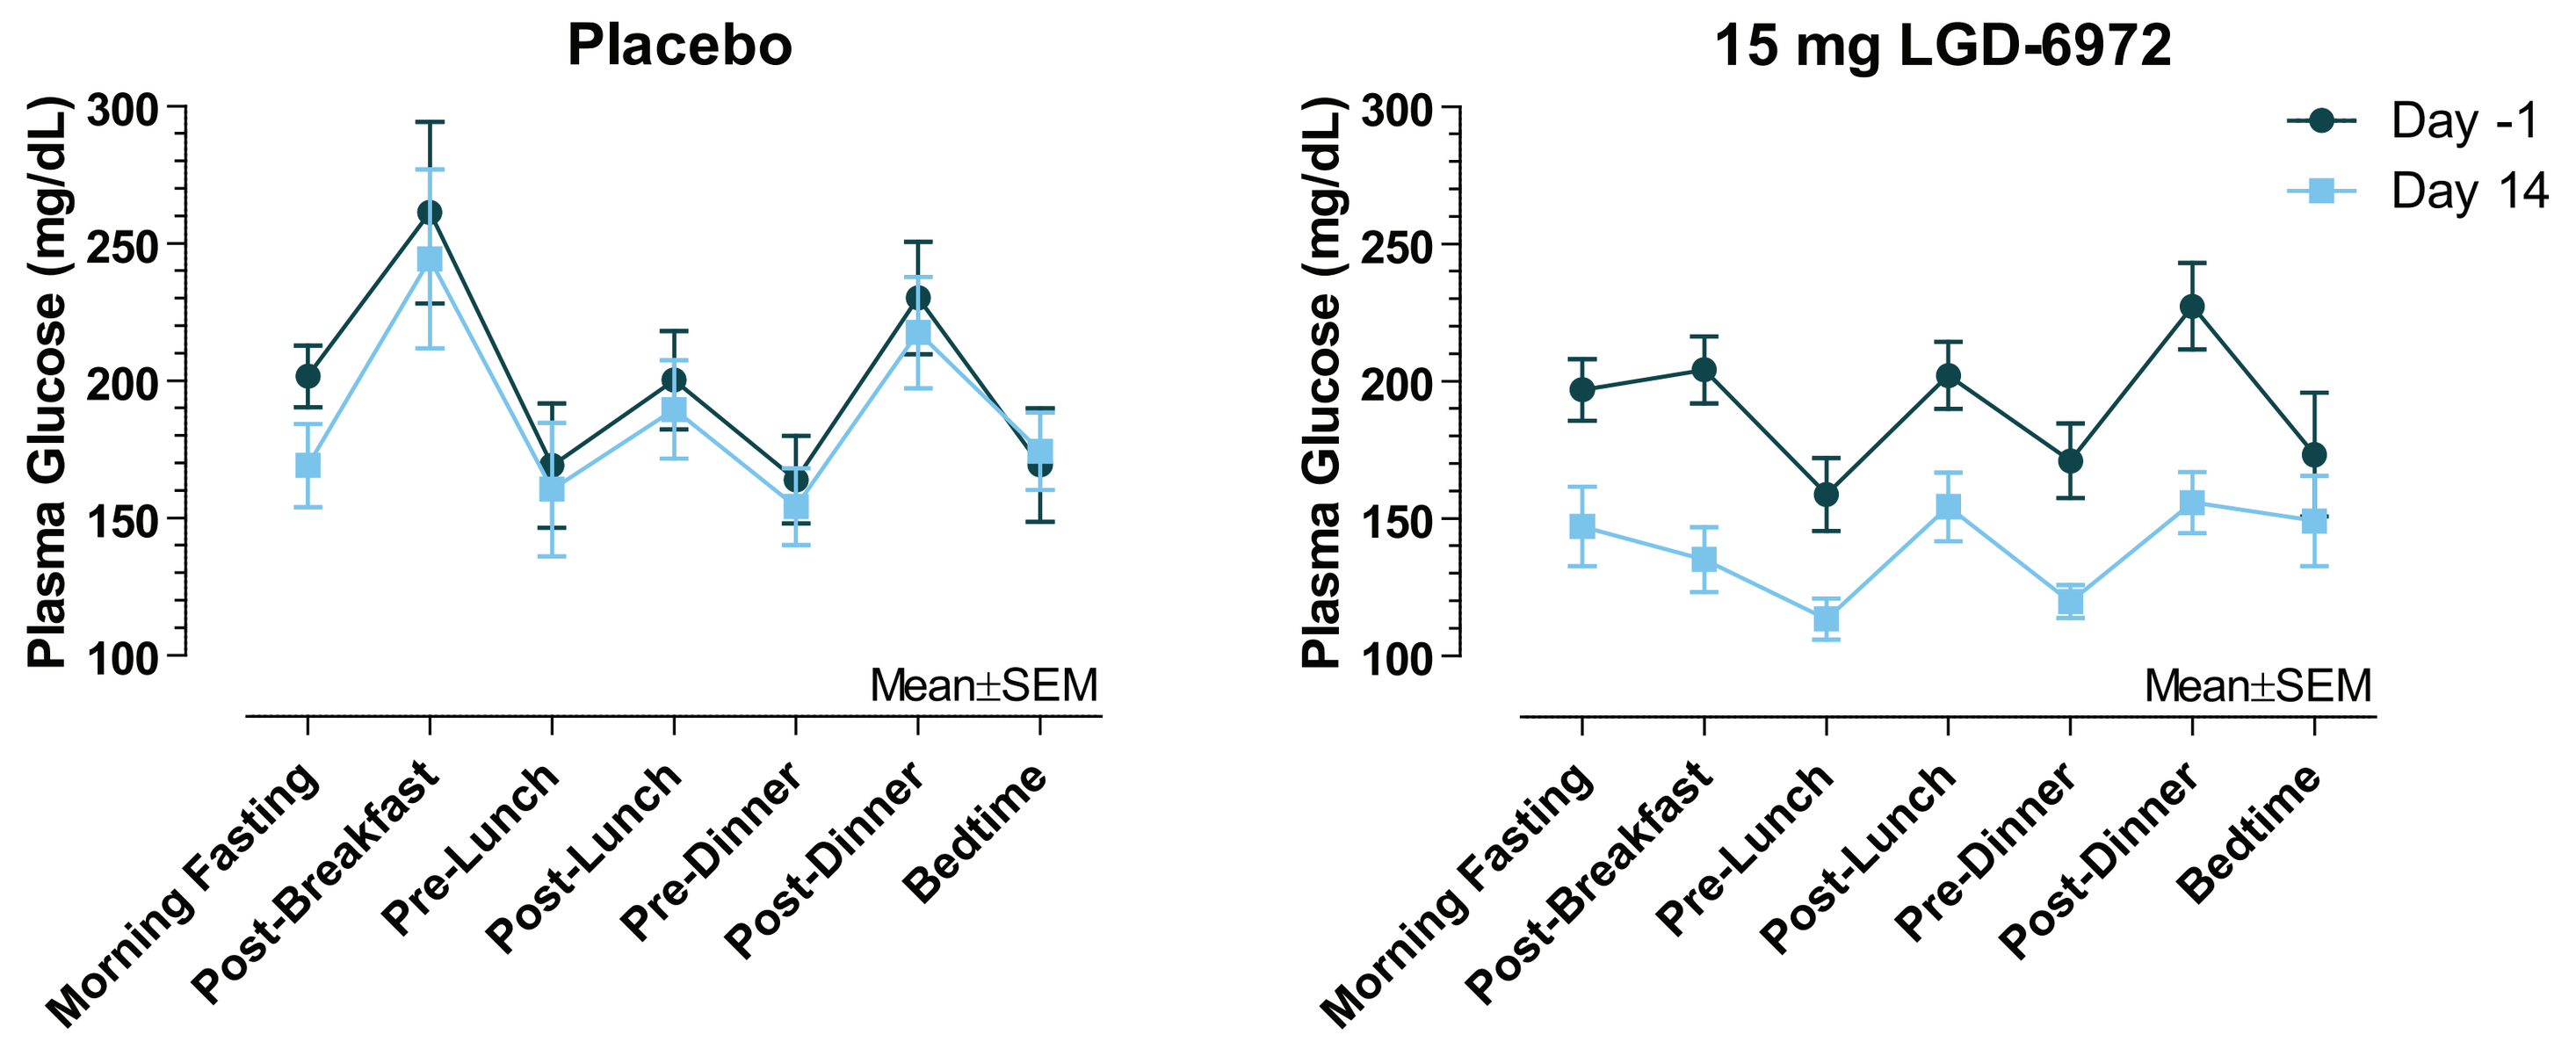
Placebo
15 mg LGD-6972
Plasma Glucose (mg/dL)
Day -1
Day 14
Mean±SEM
Morning Fasting
Post-Breakfast
Pre-Lunch
Post-Lunch
Pre-Dinner
Post-Dinner
Bedtime

Figure 4. HbA1c significantly declined from baseline in all cohorts during the course of the study. The decrease appeared greater with LGD-6972 treatment but longer term studies will be required to determine the magnitude and significance of these effects.

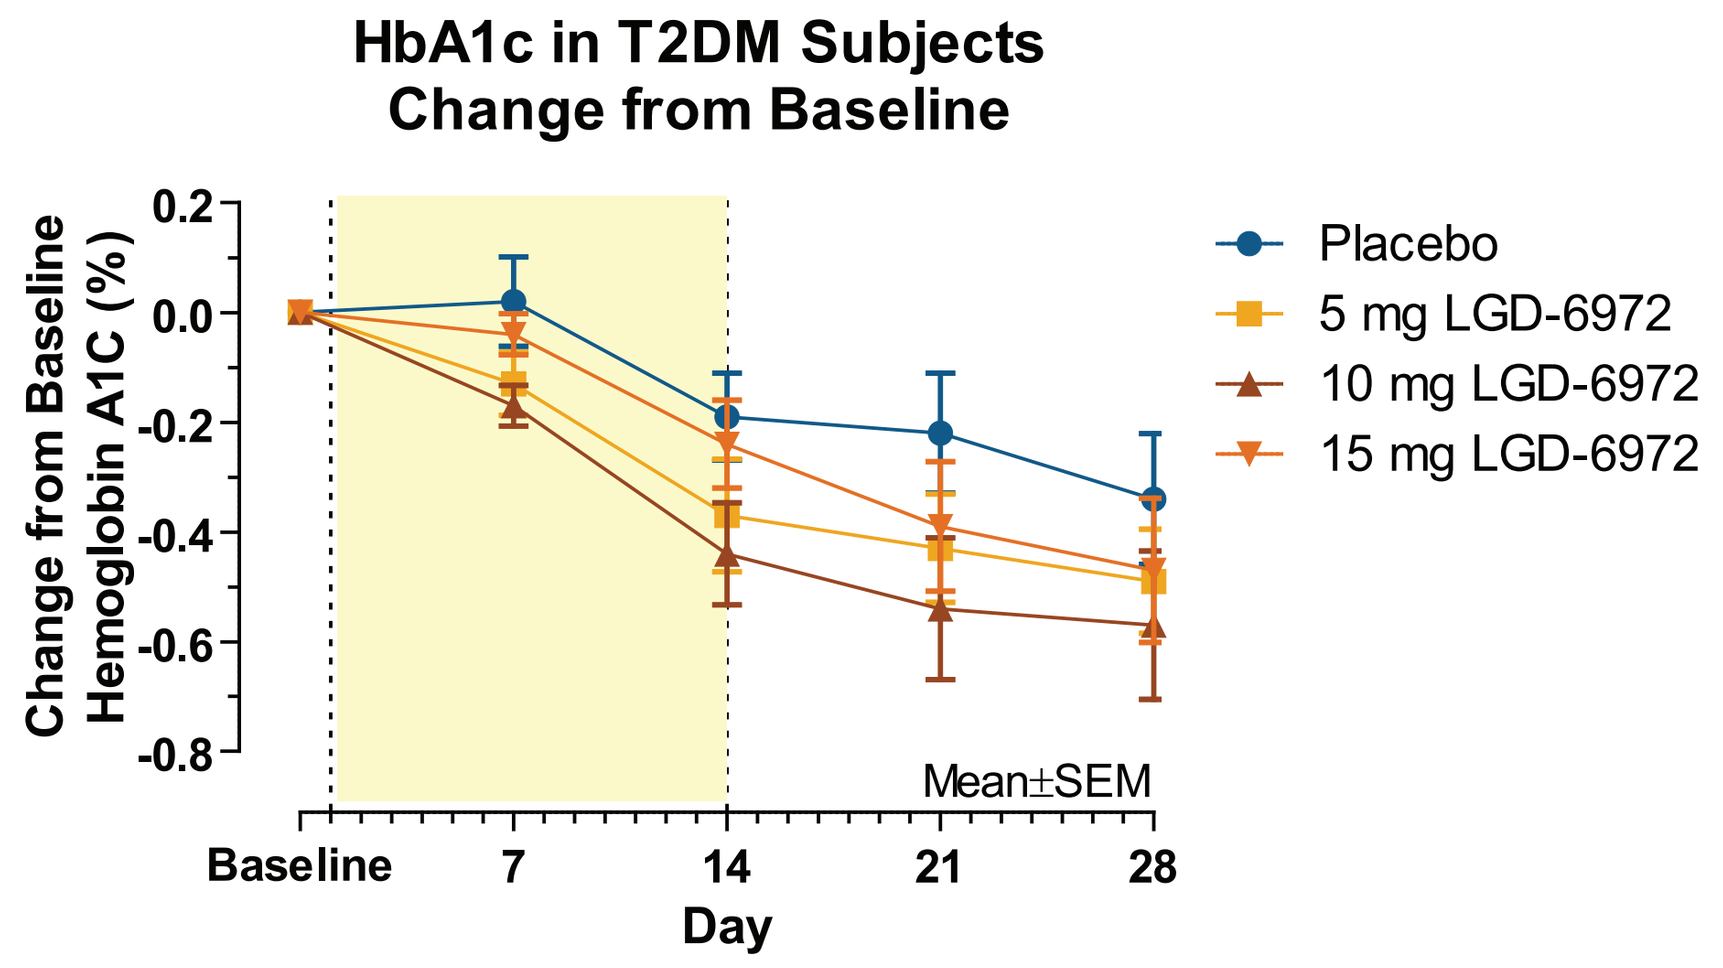
HbA1c in T2DM Subjects
Change from Baseline
Change from Baseline Hemoglobin A1C (%)
Placebo
5 mg LGD-6972
10 mg LGD-6972
15 mg LGD-6972
Mean±SEM
Day

HORMONAL EFFECTS

Figure 5. LGD-6972 increased fasting plasma glucagon and total GLP-1 in T2DM subjects in a dose dependent manner. There were no statistically significant effects on active GLP-1 concentrations.

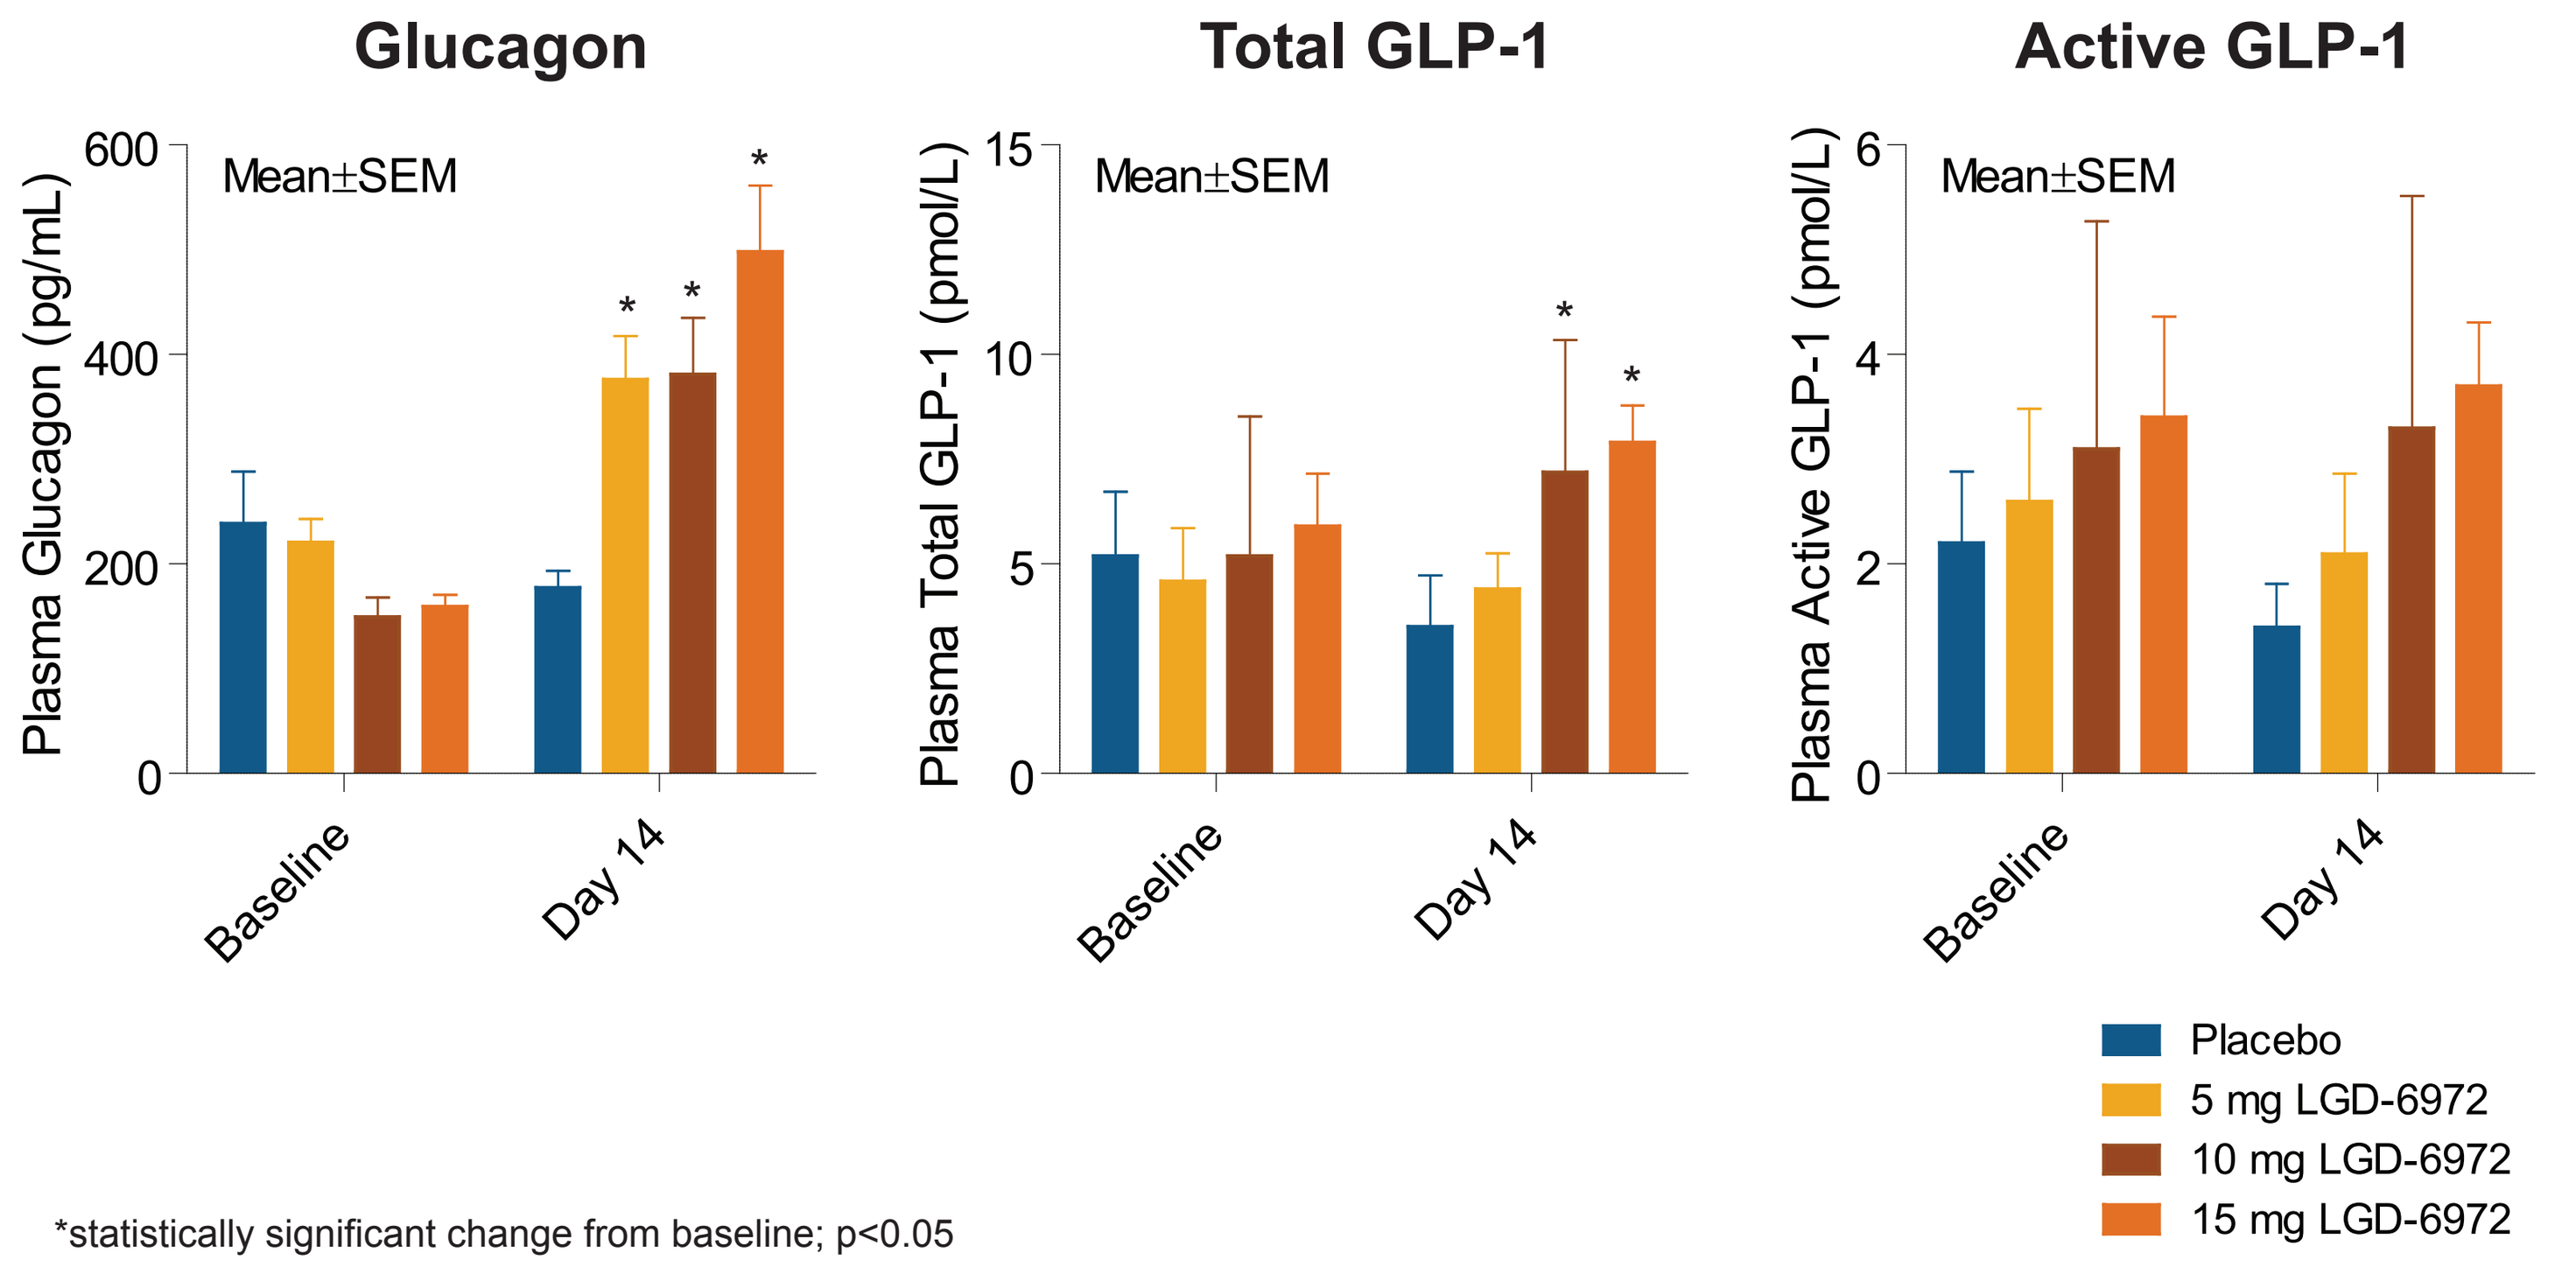
Glucagon
Total GLP-1
Active GLP-1
Plasma Glucagon (pg/mL)
Plasma Total GLP-1 (pmol/L)
Plasma Active GLP-1 (pmol/L)
Mean±SEM
Baseline
Day 14
Placebo
5 mg LGD-6972
10 mg LGD-6972
15 mg LGD-6972

*statistically significant change from baseline; p<0.05

OGTT

Figure 6. An oral glucose tolerance test was performed at baseline and on day 14 in the 10 mg cohort. There were no differences from baseline to day 14 in placebo treated subjects. Glucose was reduced on day 14 relative to baseline in the LGD-6972 treated subjects, but glucose excursion remained unchanged. In contrast, insulin excursion was increased on day 14 in LGD-6972 treated subjects. Glucagon was elevated on day 14 but showed a paradoxical decrease during the OGTT.

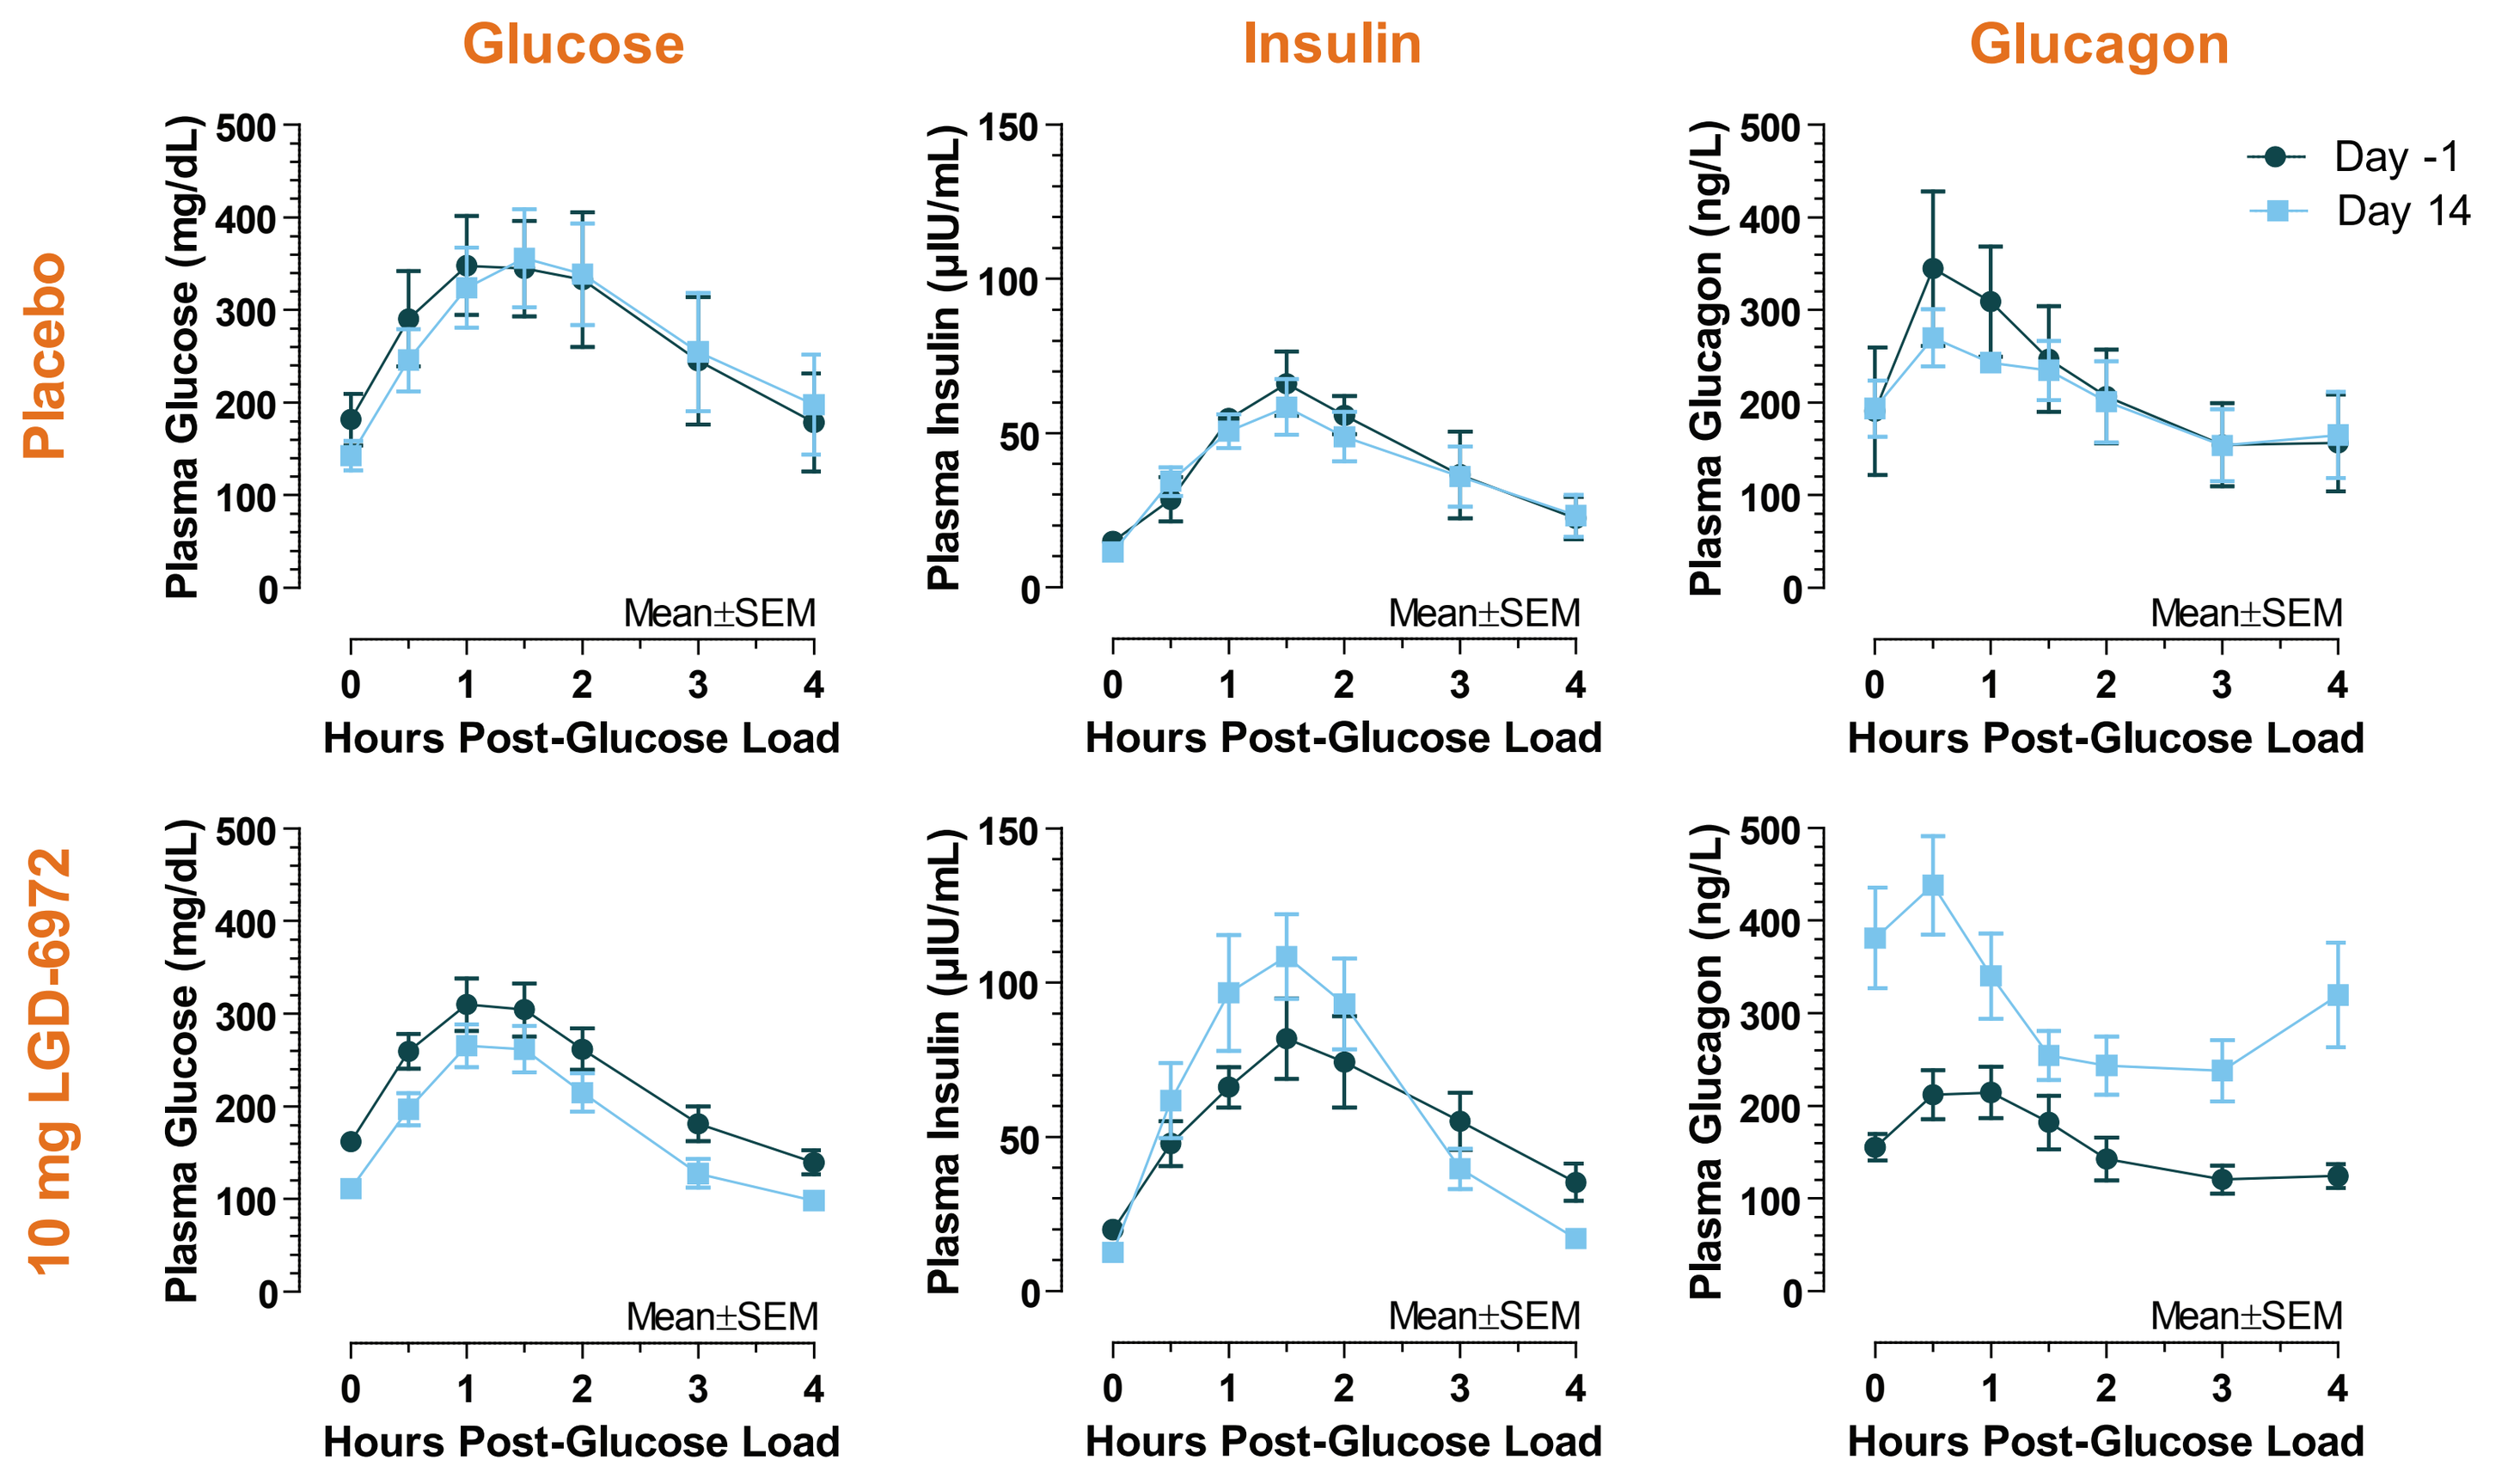
Glucose
Insulin
Glucagon
Placebo
10 mg LGD-6972
Plasma Glucose (mg/dL)
Plasma Insulin (μIU/mL)
Plasma Glucagon (ng/L)
Day -1
Day 14
Mean±SEM
Hours Post-Glucose Load

SUMMARY

- LGD-6972 is a highly effective glycemia modifying agent with a rapid onset of effect in T2DM subjects
- In a multiple ascending dose study, LGD-6972 was well tolerated in NHV and T2DM subjects
- Dose dependent decreases in fasting plasma glucose were observed in NHV and T2DM after a single dose and the effects persisted throughout a 14 day dosing period
- Glucose reductions were observed in both fasting and postprandial states
- In an OGTT, insulin excursion was increased while glucagon excursion decreased
- The safety, PK, and glycemic effects observed with 14 day administration of LGD-6972 support continued clinical development as a once daily treatment for diabetes

Levine-Riggs Symposium; Long Beach, CA, USA; March 6-9, 2016