F2016

Improved Safety of Opioid Analgesic Oliceridine Compared to Morphine Assessed by Utility Function Analysis

Albert Dahan,[1] Marieke Niesters,[1] Michael J. Fossler,[2] Mark A. Demitrack,[2] Erik Olofsen[1]

[1]Department of Anesthesiology, Leiden University Medical Center, Leiden, The Netherlands; [2]Trevena Inc., Chesterbrook, PA, USA

BACKGROUND

- Opioids produce potent pain relief and therefore remain the cornerstone of treatment of moderate to severe pain.[1]
- Among the many opioid side effects, respiratory depression may complicate adequate dose exposure for analgesia, and in rare instances can be potentially life-threatening.[2]
- In the context of the "opioid crisis", there is an ongoing search for potent opioid analgesics with reduced adverse effects.
- Full μ-receptors agonists produce analgesia, by activation of the G-coupled signaling pathway, and dose-dependent respiratory depression (with apnea at high doses) by activation of the β-arrestin pathway.[3]
- Recent focus has been on the development of biased ligands, which are μ-receptors agonists that selectively engage the G-coupled signaling pathway while avoiding the β-arrestin pathway[3,4] **(Figure 1)**.

Figure 1: New Hypothesis: GPCRs Have Distinct Signaling Pathways

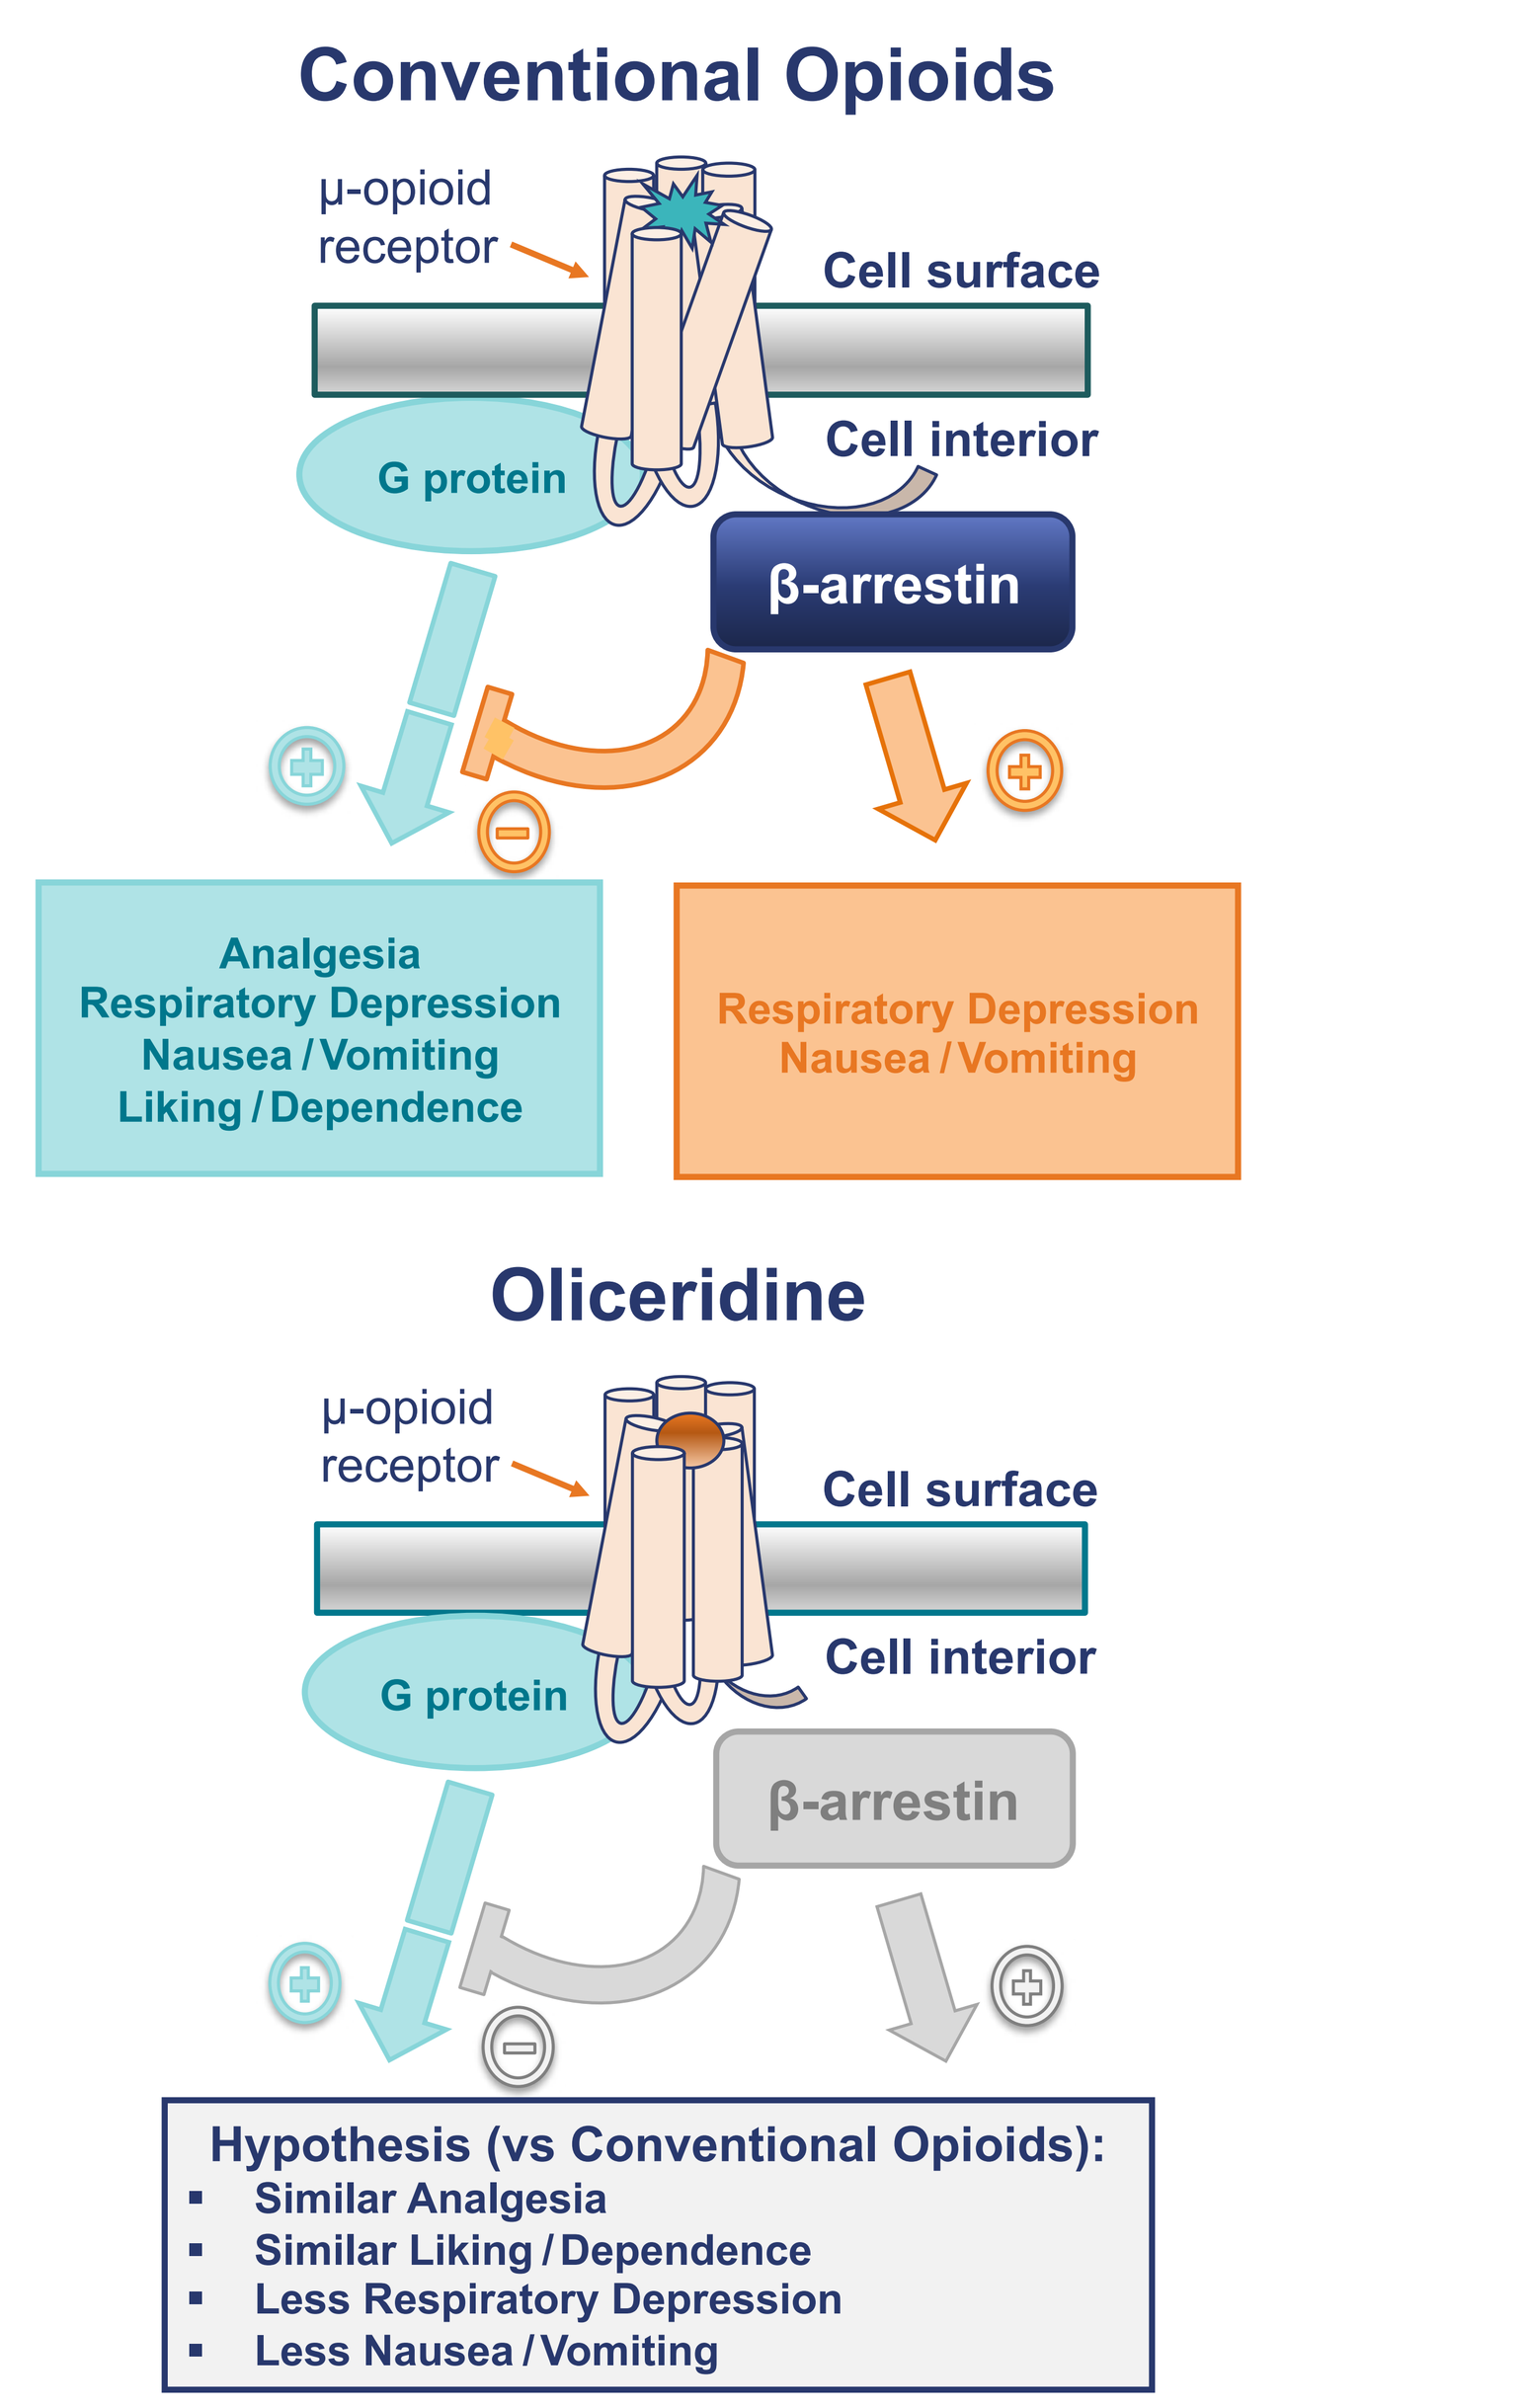
Conventional Opioids
μ-opioid receptor
Cell surface
Cell interior
G protein
β-arrestin
Analgesia
Respiratory Depression
Nausea / Vomiting
Liking / Dependence
Respiratory Depression
Nausea / Vomiting
Oliceridine
μ-opioid receptor
Cell surface
Cell interior
G protein
β-arrestin
Hypothesis (vs Conventional Opioids):
Similar Analgesia
Similar Liking / Dependence
Less Respiratory Depression
Less Nausea / Vomiting

- In a previously published study of healthy volunteers, oliceridine (TRV130) at doses of 1.5 mg, 3 mg, and 4.5 mg elicited rapid analgesic effect, with improved analgesia at the higher doses of 3 and 4.5 mg compared to morphine 10 mg[5] (Figure 2).
- In this study, oliceridine, also exhibited less reduction in respiratory drive compared to morphine, as measured by the ratio of minute ventilation over end-tidal CO_2 on ventilatory response to hypercapnia (VRH) testing[5] **(Figure 2)**.

Figure 2: Analgesic effect and VRH response of Oliceridine

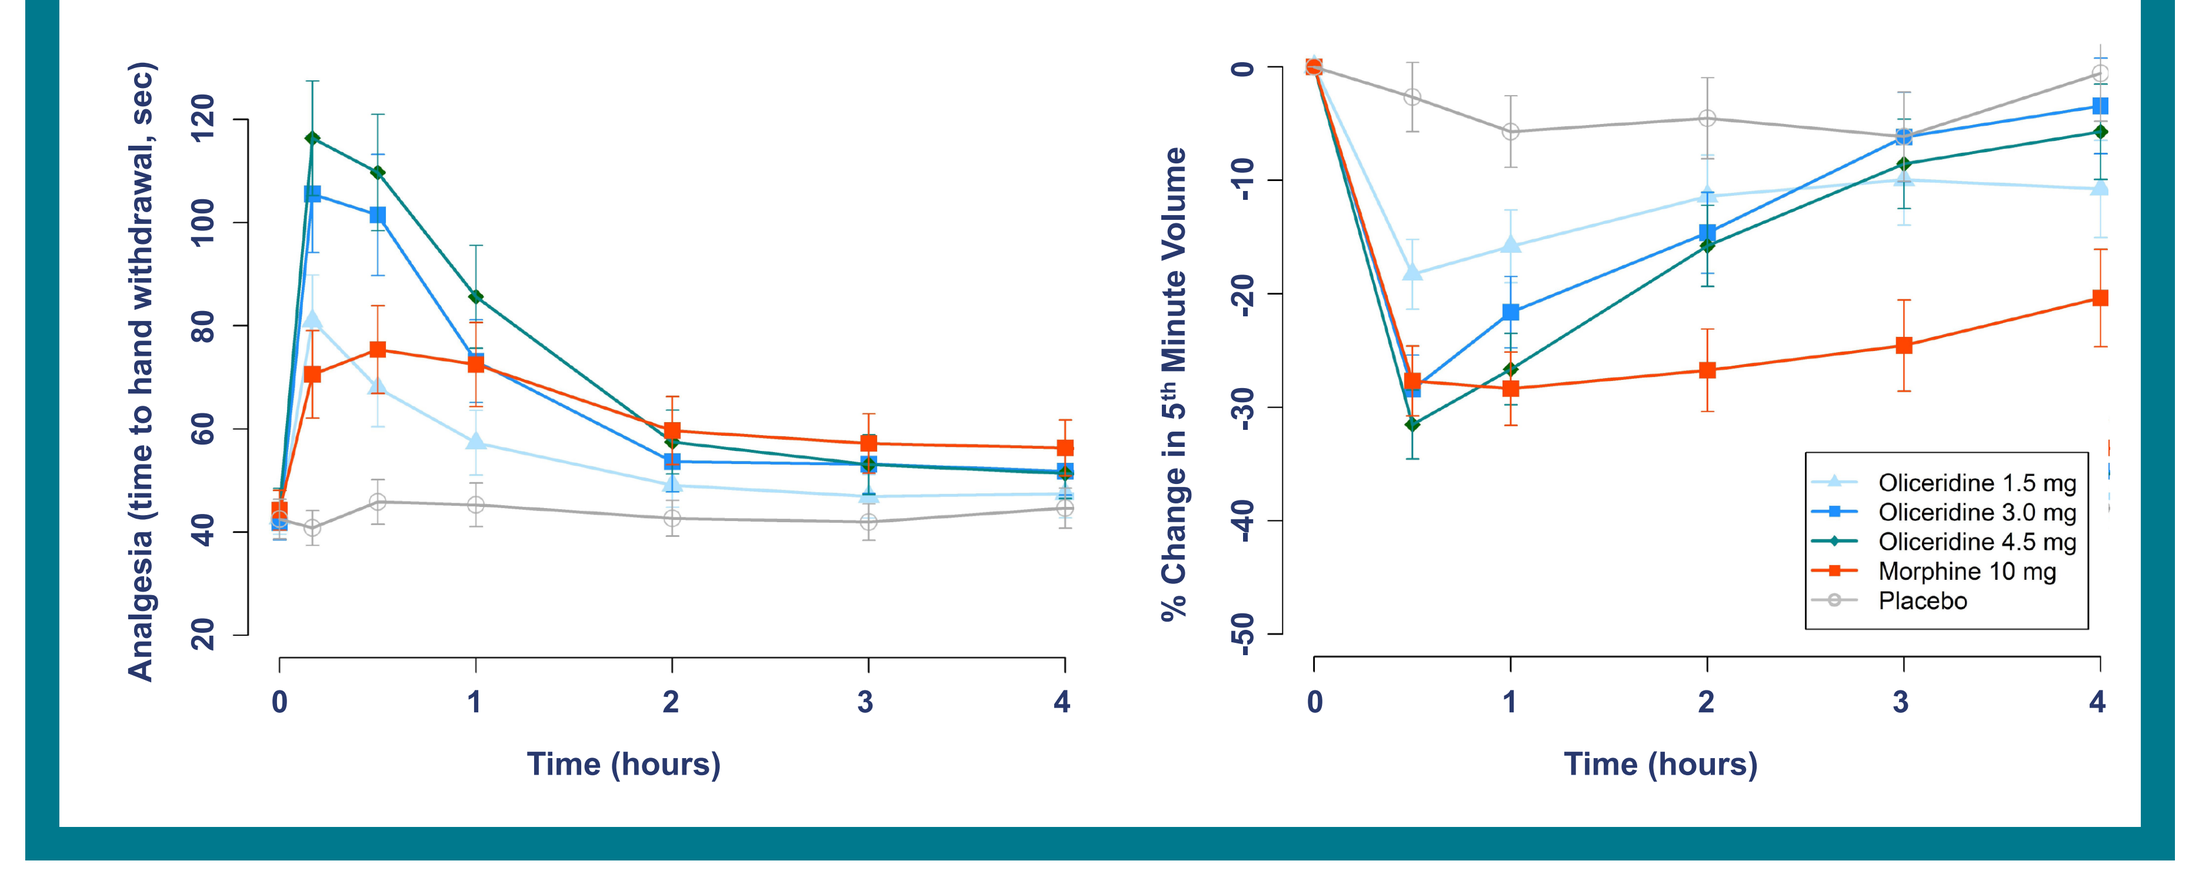
Analgesia (time to hand withdrawal, sec)
Time (hours)
% Change in 5th Minute Volume
Time (hours)
Oliceridine 1.5 mg
Oliceridine 3.0 mg
Oliceridine 4.5 mg
Morphine 10 mg
Placebo

OBJECTIVE

- Here we reanalyzed data from the above study and performed a population pharmacokinetic-pharmacodynamic (PKPD) analysis, which allowed us to construct safety or utility functions U, which give an integrated profile on the probability of drug harm in the light of its benefit.

METHODS

- We performed a PK/PD analysis that gives parameter estimates + an indication of inter-individual variabilities (ω^2)
- The population PKPD analyses were performed in NONMEM.
- Utility functions are objective and precise assessments of the probability of analgesia relative to the probability of respiratory depression (R).
- We created the classical utility function (benefit minus harm):
 - U1 = P(A) – P(R) ; where A is analgesia and R is respiratory depression.
 - and a novel function (benefit without harm or the probability of analgesia without respiratory depression); U2 = P(A AND NOT R).
- Taking the parameter estimates and their varibility into account, we performed 2 x 10,000 simulations for analgesia (A) and respiratory depression (R).
- We determined the probability of the occurence of analgesia (A) and of respiratory depression (R), based on pre-determined thresholds.
- Thresholds are for example P(A > 50%) or P (R < 50%). The utility is then P(A > 0.5) − P(R > 0.5); this is the classical utility function.
- A new utility function, A without R, was also calculated: P(A > 0.5 AND R < 0.5).

RESULTS

PK/PD analysis

- The morphine steady-state plasma concentration causing 25% RD was 11 ± 2 ng/mL (median ± SEM) and for concentration causing a doubling of the pain tolerance 34 ± 10 ng/mL.
- The equivalent values for oliceridine were 27 ± 4 ng/mL (ventilation) and 28 ± 5 ng/mL.
- **These values are indicative of a 2.5-fold greater morphine respiratory potency compared to olicerdine while equipotency was observed for the analgesic efficacy of the two opioids.**
- Additionally, oliceridine equilibrates more rapidly than morphine within its effect compartment.

Utility Functions

- The two utility curves that were constructed, i.e. the probability of analgesia minus the probability of RD and the probability of analgesia without RD, were all in favor of oliceridine compared to morphine **(Figure 3 and Figure 4).**
- This indicates that **following treatment with oliceridine the probability of analgesia exceeds that of RD**, over the dose range studied, in contrast to morphine, where the probability of RD exceeded that of analgesia.

Figure 3: Utility Curves Oliceridine vs Morphine

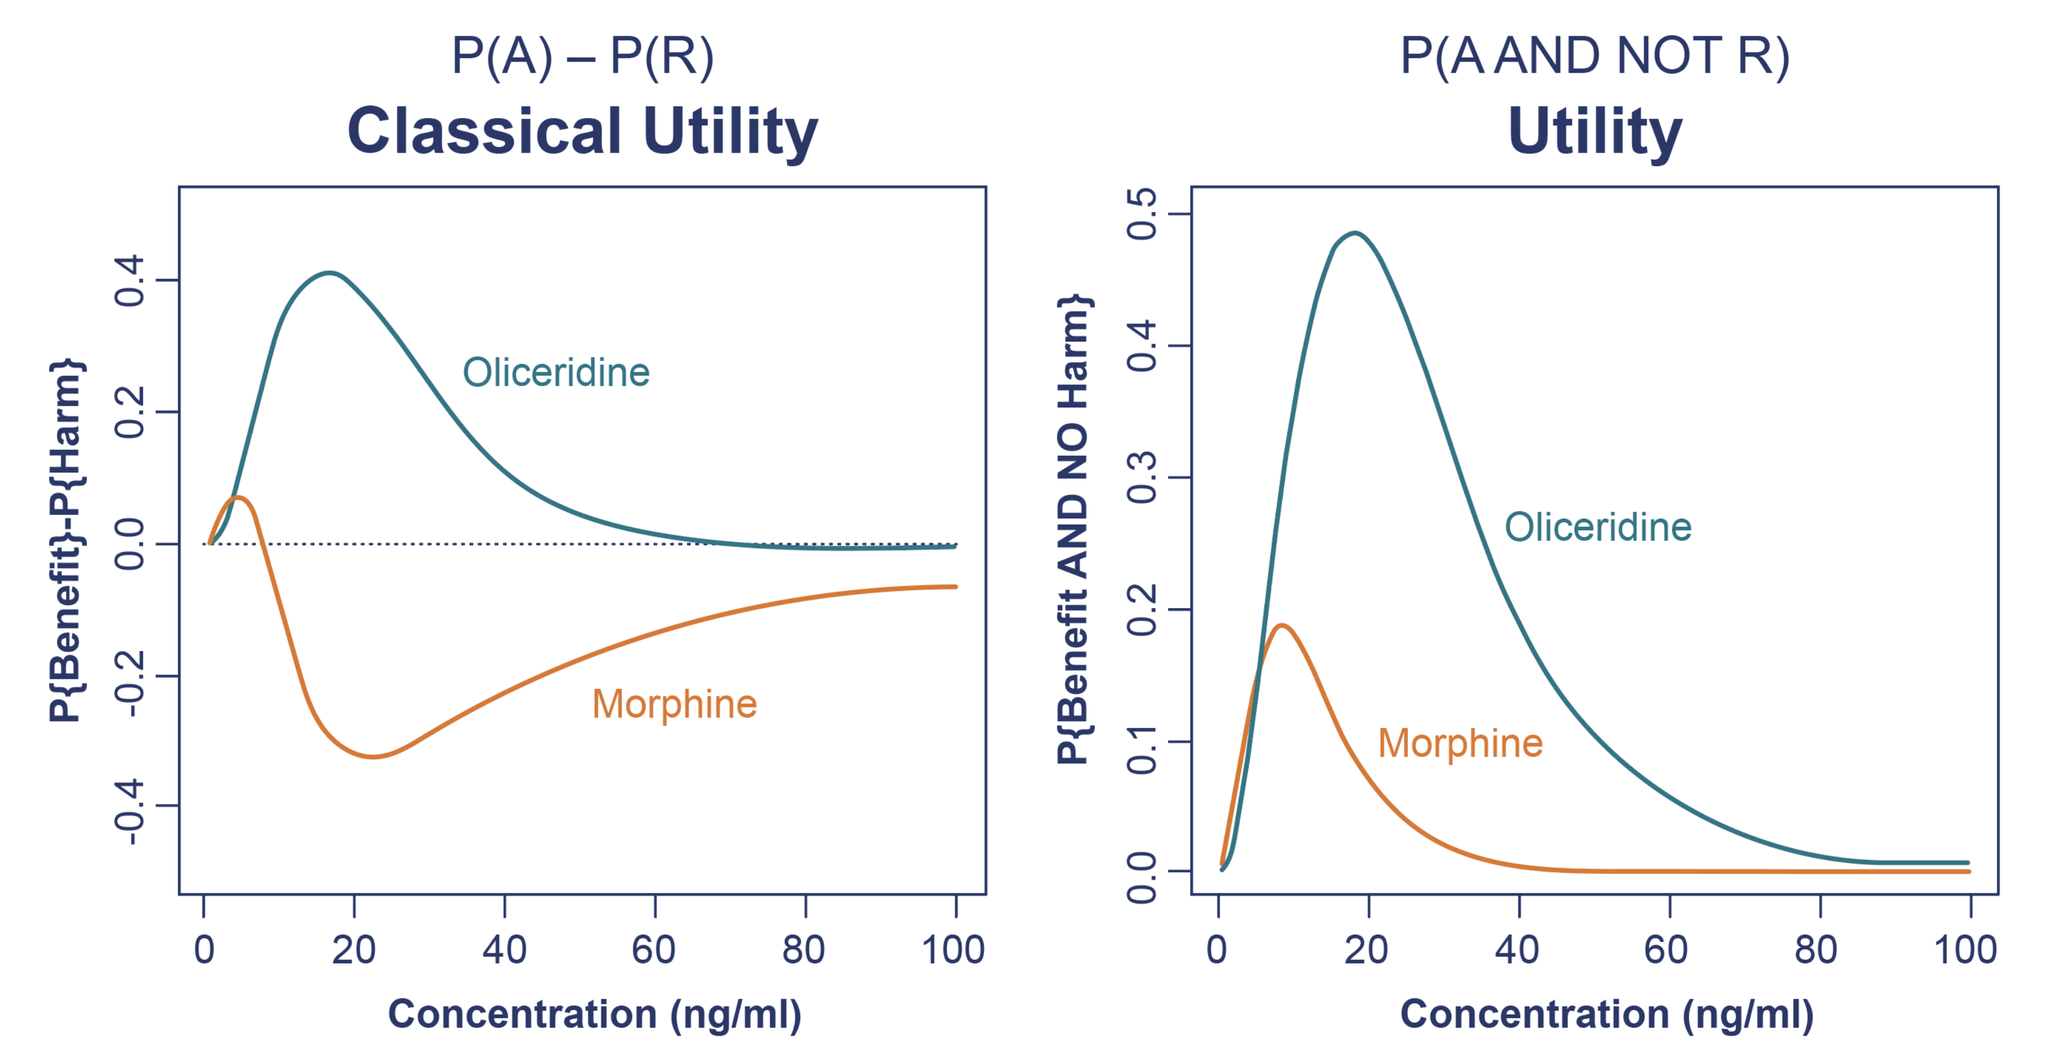
P(A) – P(R)
Classical Utility
P{Benefit}-P{Harm}
Oliceridine
Morphine
Concentration (ng/ml)
P(A AND NOT R)
Utility
P{Benefit AND NO Harm}
Oliceridine
Morphine
Concentration (ng/ml)

Figure 4A: MORPHINE Classic and Novel Utility Functions

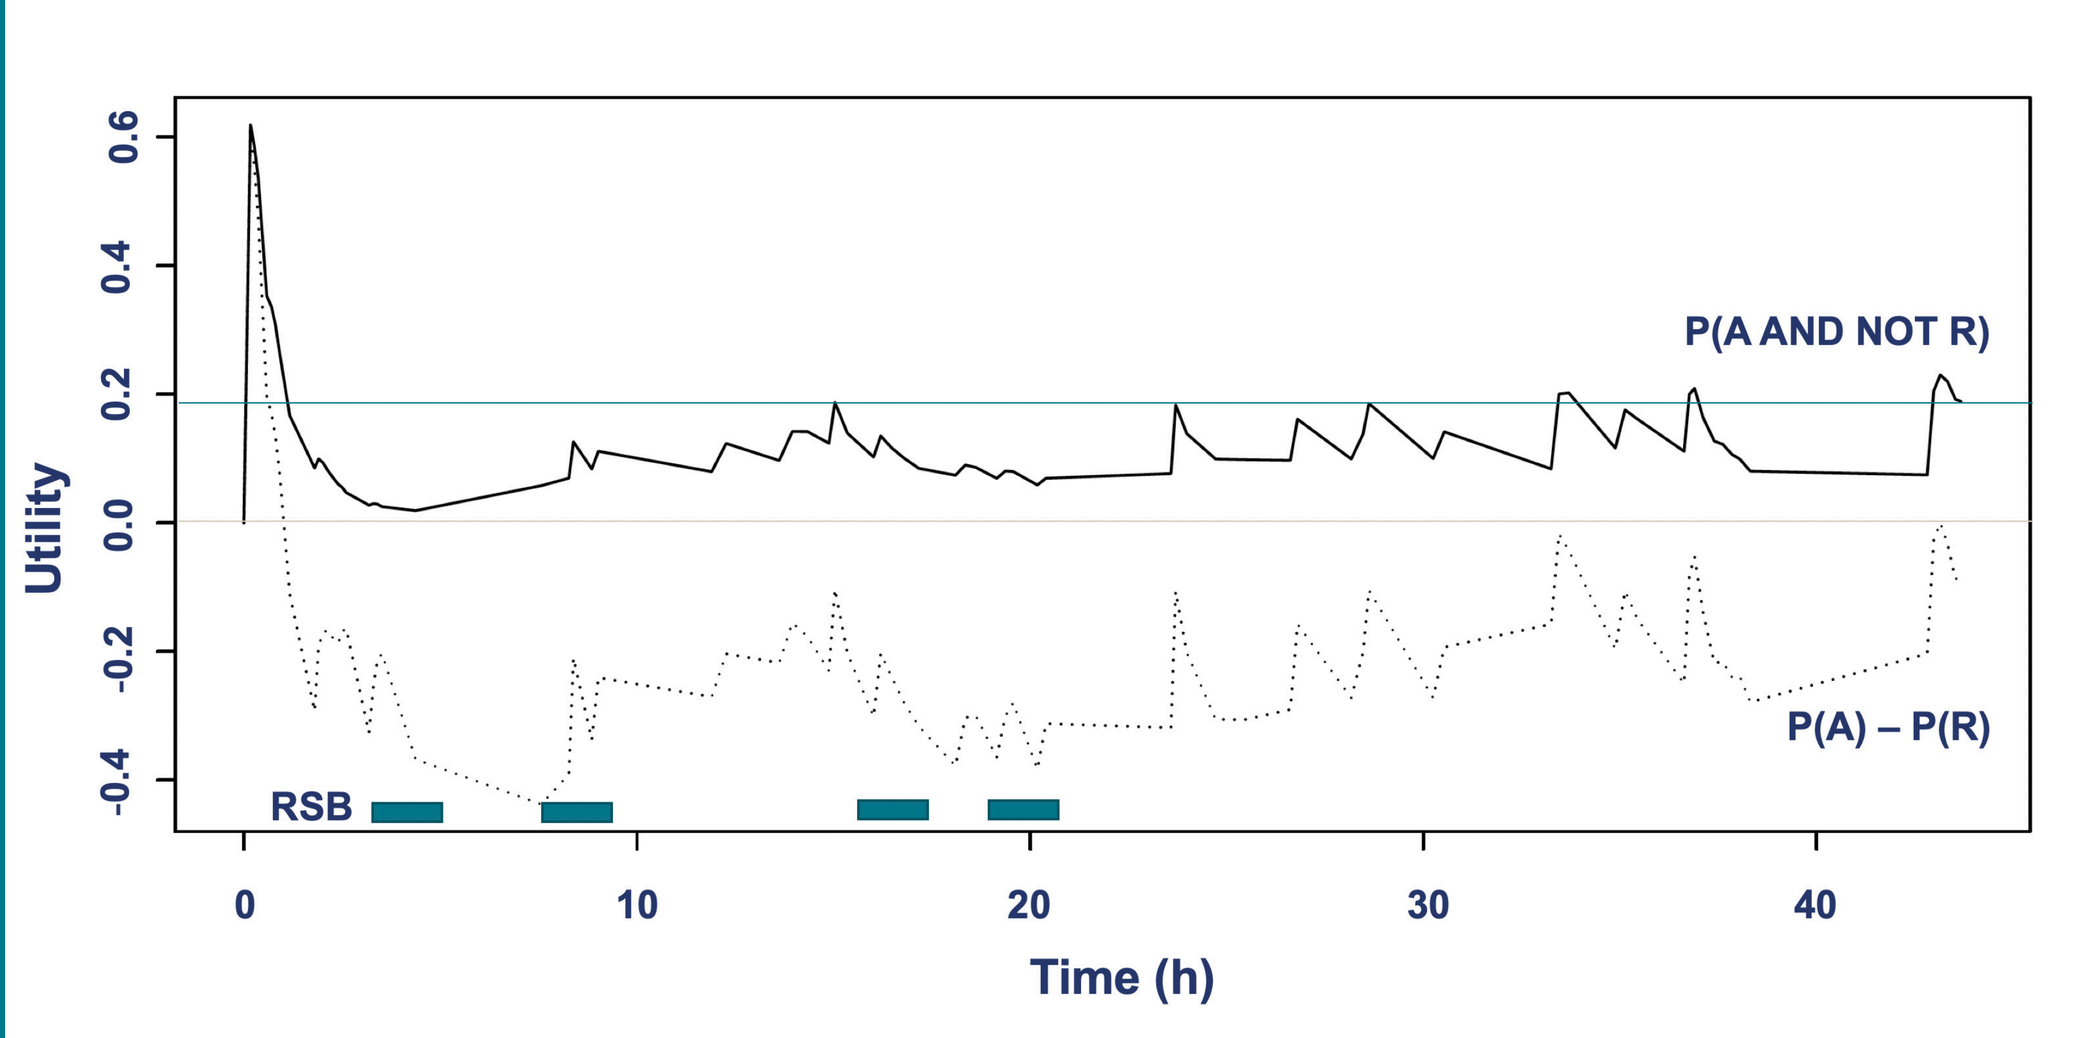
Utility
P(A AND NOT R)
P(A) – P(R)
RSB
Time (h)

Figure 4B: OLICERIDINE Classic and Novel Utility Functions

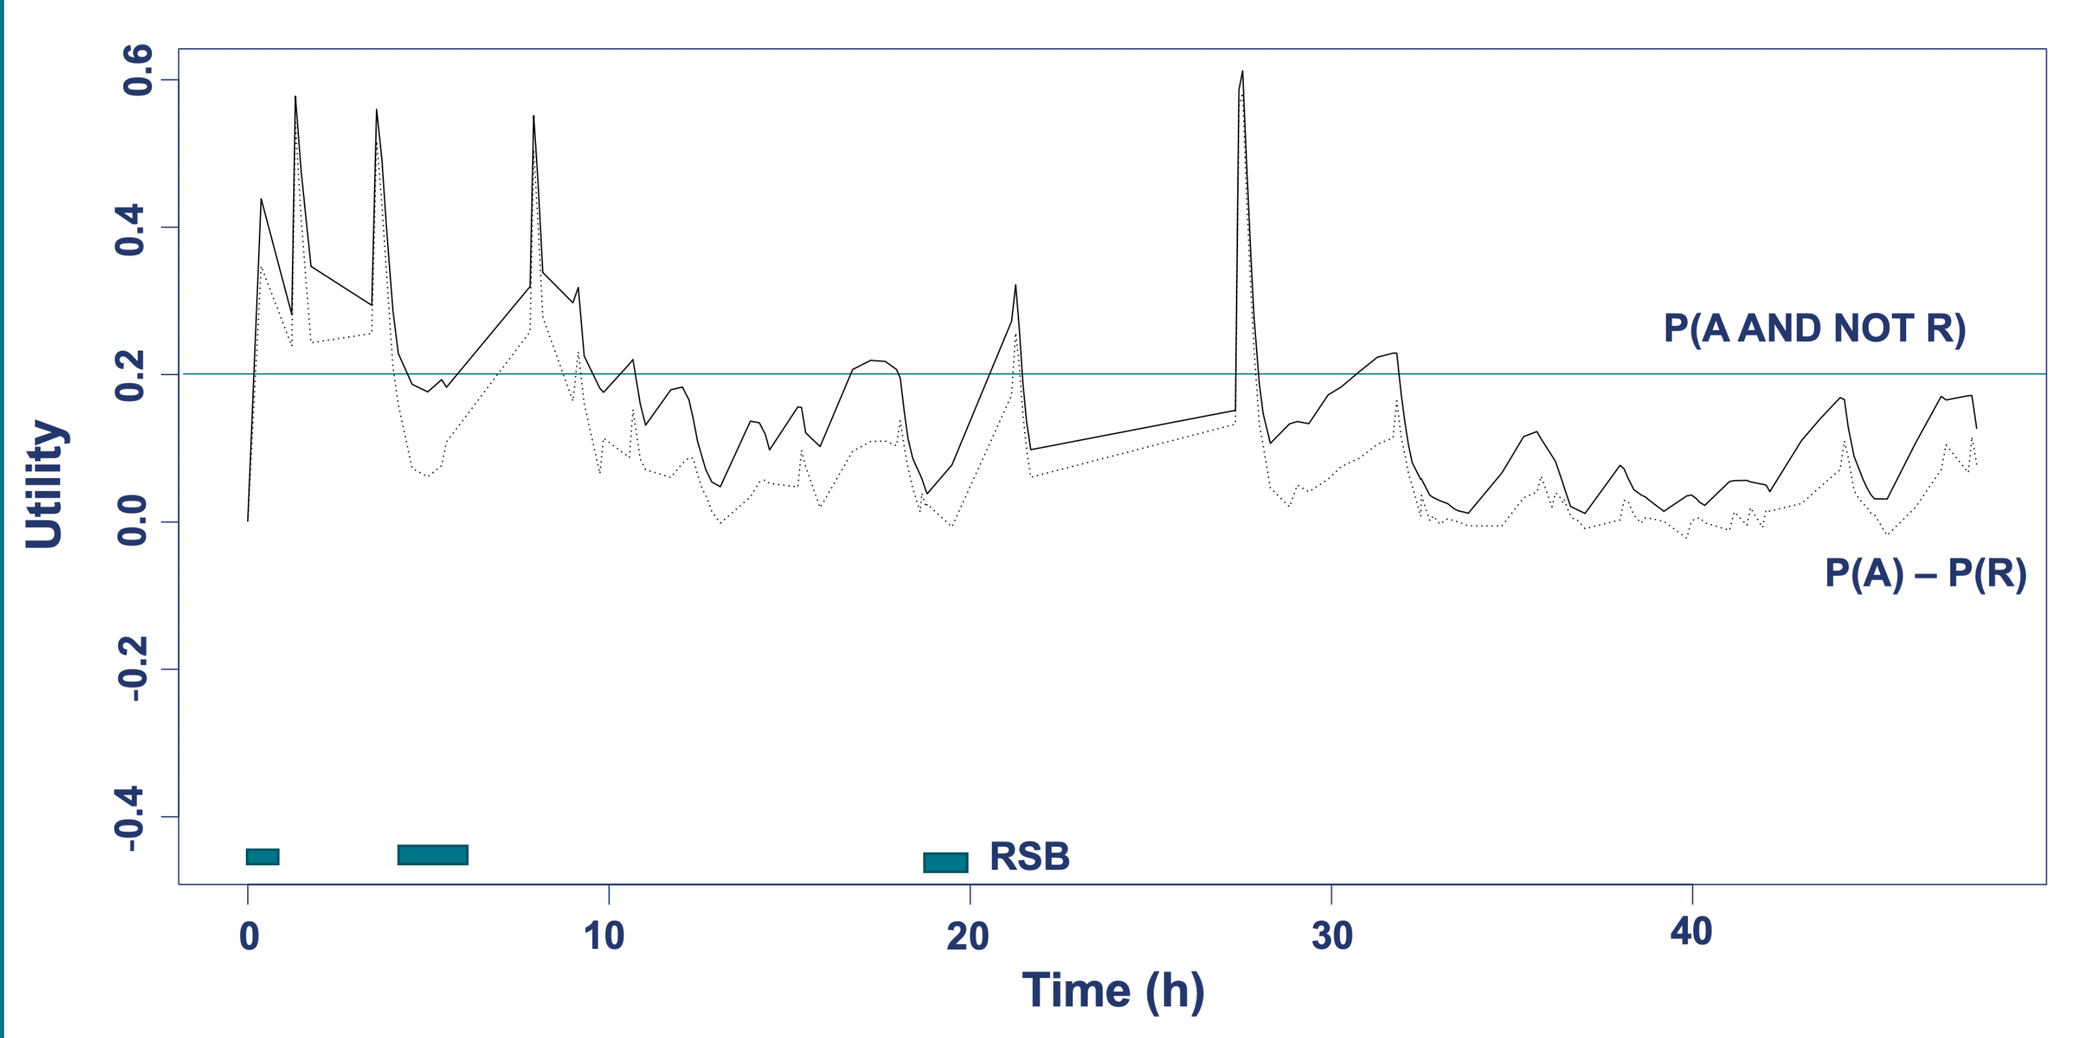
Utility
P(A AND NOT R)
P(A) – P(R)
RSB
Time (h)

CONCLUSION

- The PK-PD analysis revealed a more than 2-fold greater potency of morphine compared to oliceridine in producing respiratory depression at doses where the two opioids were equianalgesic.
- Oliceridine has a greater analgesic probability combined with a lesser probability of respiratory depression.
- The clinical respiratory events occurred in clinical practice when the utility P(A NOT R) < 0.2. The experimental utility may well be translated into clinical practice.
- Compared to the prototypical opioid analgesia morphine, the G-protein selective μ agonist, oliceridine, has a favorable safety profile when considering both analgesia and respiratory depression.

Reference:

1. Shafi S, et al. JAMA surgery. 2018;153(8):757-763.
2. Overdyk F,et al. Pain Manag. 2014;4(4):317-325.
3. DeWire SM, et al.J Pharmacol Exp Ther. 2013;344(3):708-717.
4. Violin JD, et al. Trends in pharmacological sciences. 2014;35(7):308-316.
5. Soergel DG, et al. Pain. 2014;155(9):1829-1835.

Disclosure and Acknowledgements:

Oliceridine is an investigational IV opioid not approved by the FDA. Layout of the poster provided by Jenna Burnip from LARK & CO. CREATIVE, and funded by Trevena, Inc.

Presented at the 2019 Annual Meeting of the American Society of Anesthesiologists (ASA 2019); October 19-23, Orlando, FL