REL-1017 (esmethadone) DID NOT PRODUCE INITIAL OR CUMULATIVE NEUROTOXIC EFFECTS OR OTHER EVIDENCE OF DAMAGE TO CORTICAL NEURONS

Marco Pappagallo [1,5], Francesco Bifari [2], Michael Bleavins [3], Franco Folli [4], Paolo Manfredi[5]

[1] Department of Anesthesiology, Albert Einstein College of Medicine, Bronx, New York, NY, USA; [2] Department of Medical Biotechnology and Translational Medicine, University of Milan, Milan, Italy; [3] University of Michigan School of Public Health, Ann Arbor, MI, USA; [4] University of Milano School of Medicine, Milan, Italy; [5] Relmada Therapeutics, Inc., New York, NY, USA

BACKGROUND

Olney's lesions Definition

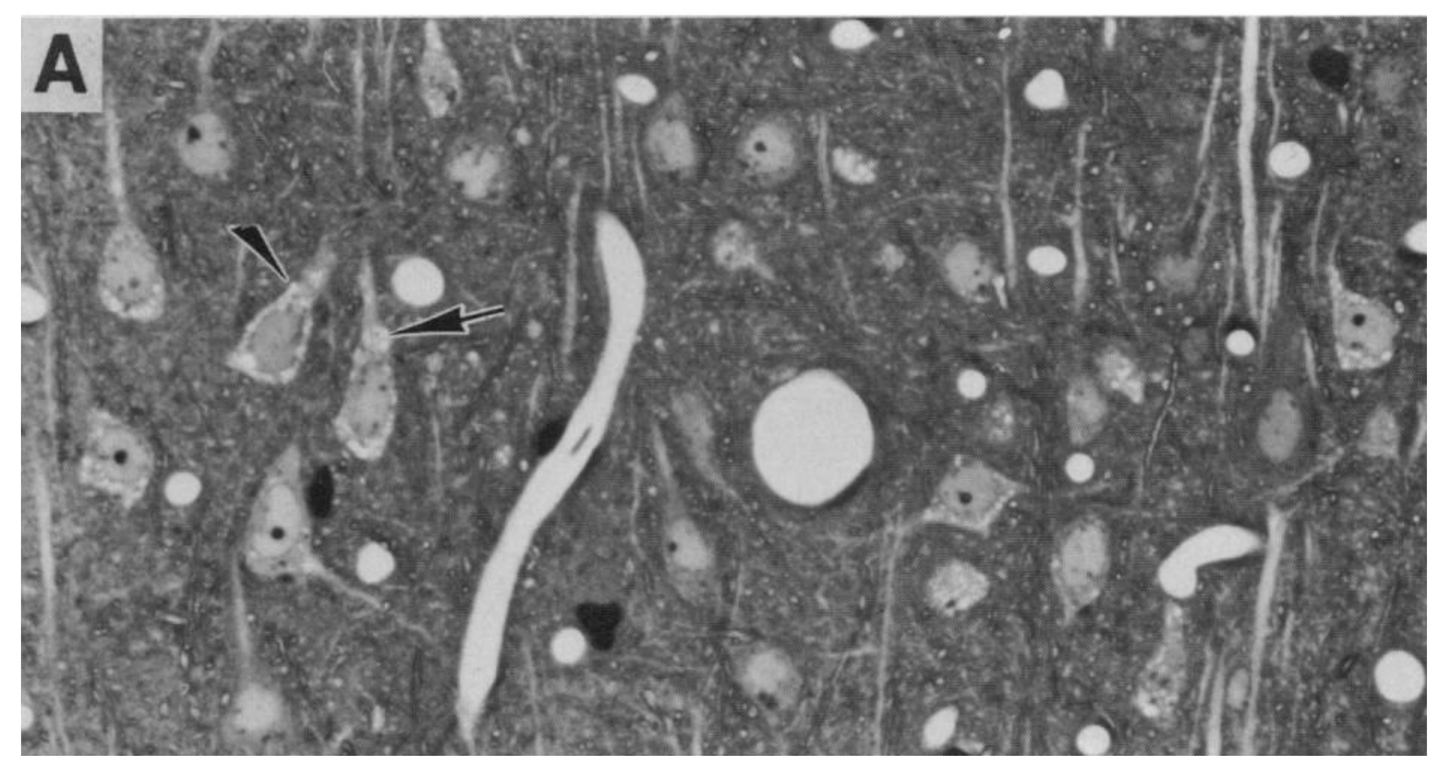
A

from J. W. Olney *Science,* Vol. 254, No. 5037 (1991)

Olney's lesions are characteristic vacuolizations (feature of neurotoxicity and potentially irreversible damage in the CNS), which can be seen by light microscopy in neurons of rats, mainly in posterior cingulate and retrosplenial cortices, after administration of select ***NMDAR channel blockers***, like MK-801, ketamine and dextromethorphan[1].

REL-1017 (esmethadone) is a promising NMDAR channel blocker

Test item IC_{50} (μM)	NMDAR type			
	hGluN1-hGluN2A	hGluN1-hGluN2B	hGluN1-hGluN2C	hGluN1-hGluN2D
REL-1017 esmethadone IC_{50} (μM)	49	28	21	69
Memantine IC_{50} (μM)	35	8.8	3.3	6.9
(±)-Ketamine IC_{50} (μM)	27	6.3	3.4	11
(+)-MK-801 IC_{50} (μM)	0.28	0.061	0.57	0.78
Dextromethorphan IC_{50} (μM)	51	15	5.8	27

REL-1017(esmethadone) is the d-optical isomer of the racemic mixture d-l-methadone. REL-1017 has been shown to have robust, rapid and sustained antidepressant effects in humans (Relmada data on file). Esmethadone is free from clinically relevant opioid effects and free from dissociative effects seen with other NMDAR channel blockers. Esmethadone has been shown to exert antidepressant-like effects in all tested murine models of depression[2].

Esmethadone's estimated potency at NMDAR subtypes in relation to FDA approved NMDAR channel blockers and MK-801 is shown in the table above (Relmada data on file)

REL-1017 (esmethadone) is a promising low affinity, low potency NMDAR channel blocker, presently undergoing Phase 3 clinical trials for the treatment of major depressive disorder (MDD).

STUDY DESIGN

Group randomization

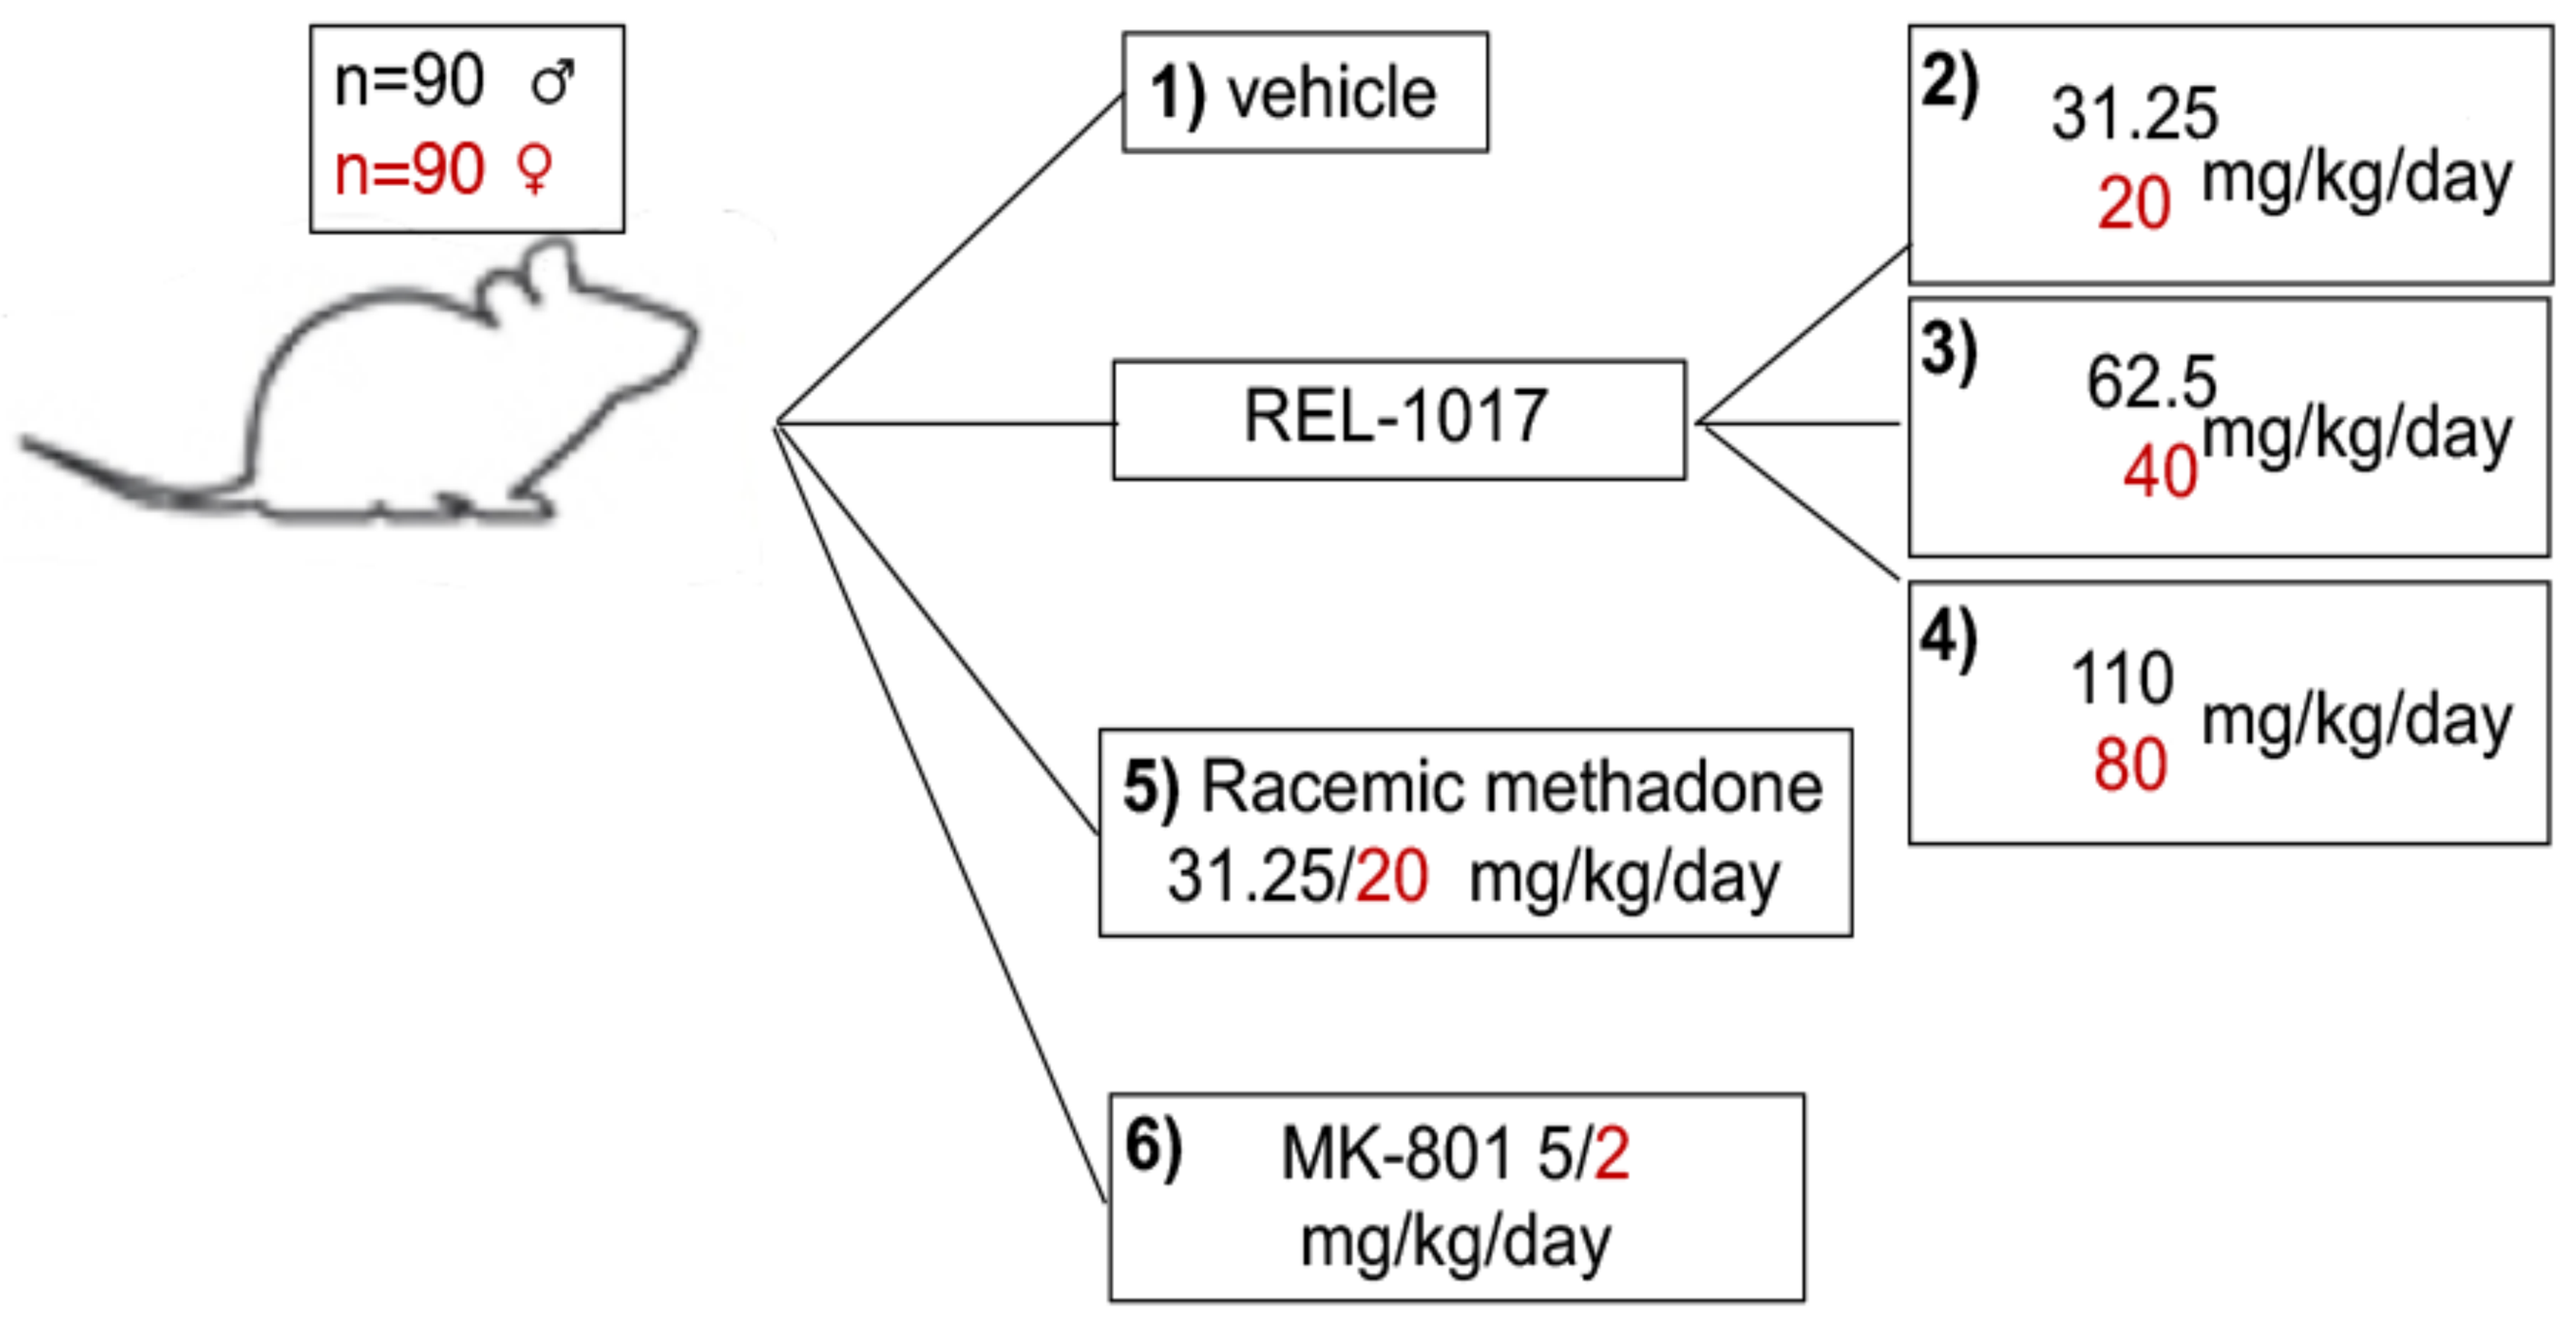
n=90 ♂
n=90 ♀
1) vehicle
REL-1017
2) 31.25 20 mg/kg/day
3) 62.5 40 mg/kg/day
4) 110 80 mg/kg/day
5) Racemic methadone 31.25/20 mg/kg/day
6) MK-801 5/2 mg/kg/day

Experimental design

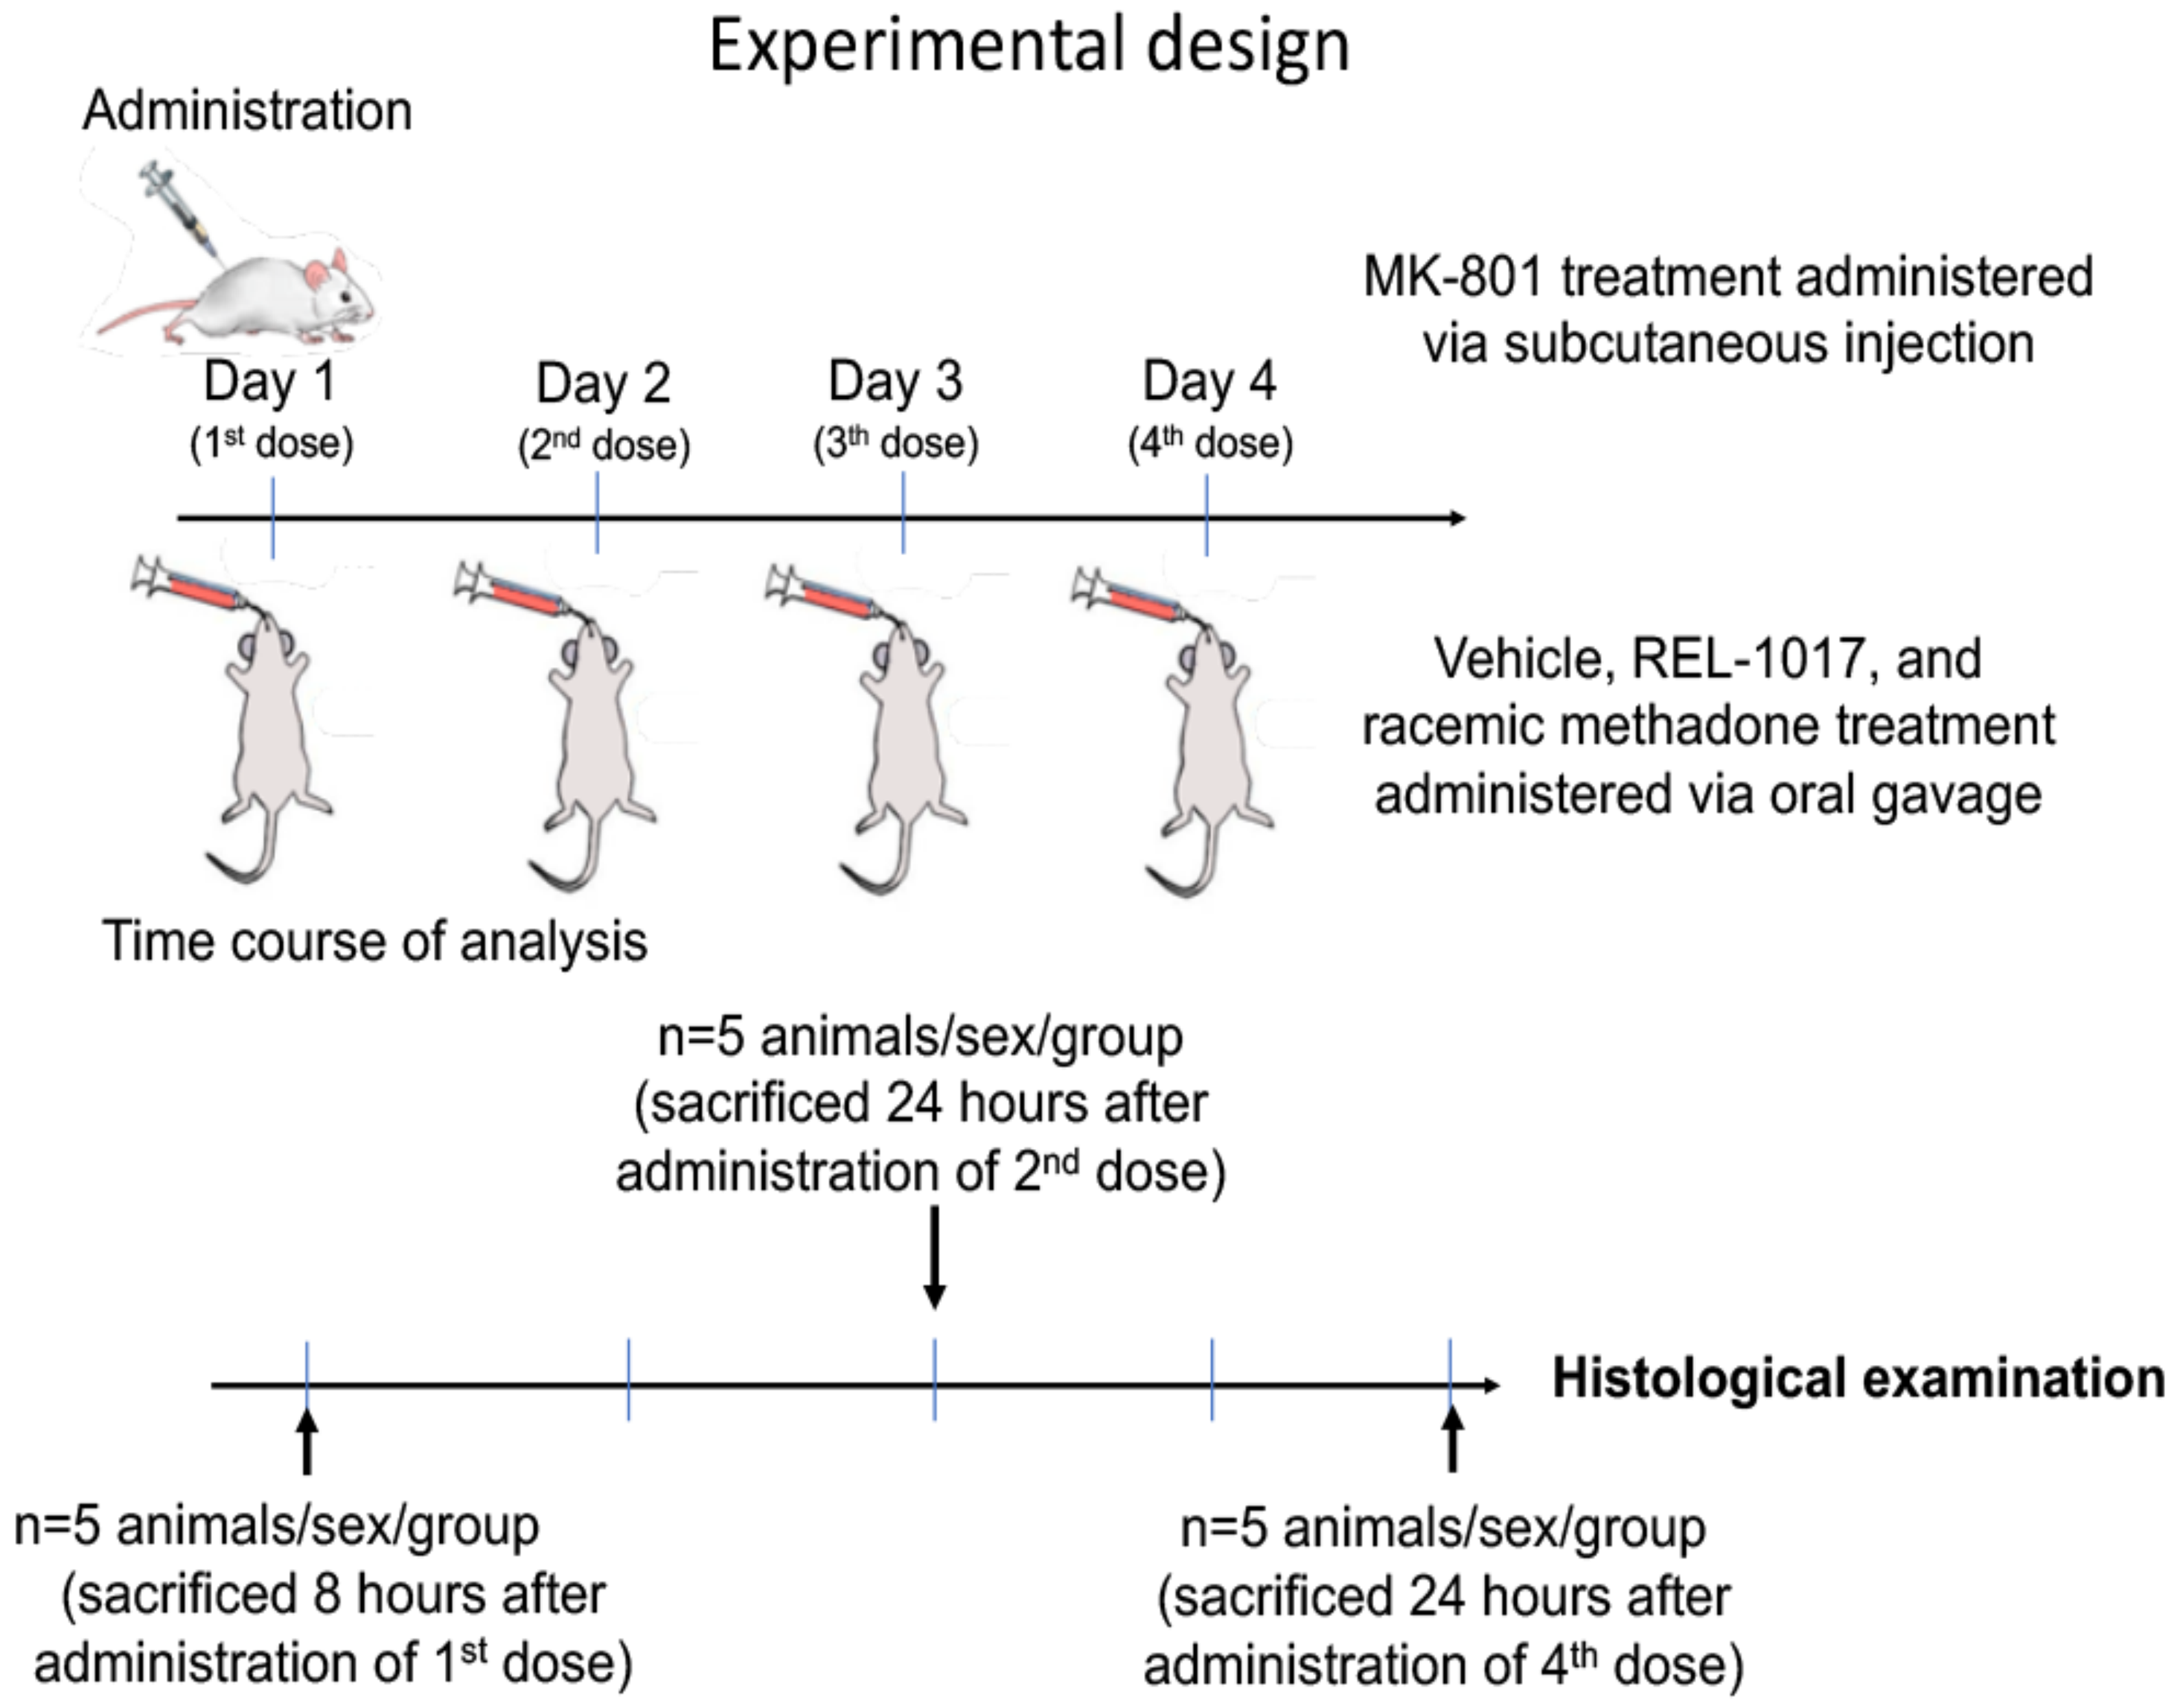
Administration
Day 1 (1st dose)
Day 2 (2nd dose)
Day 3 (3th dose)
Day 4 (4th dose)
MK-801 treatment administered via subcutaneous injection
Vehicle, REL-1017, and racemic methadone treatment administered via oral gavage
Time course of analysis
n=5 animals/sex/group (sacrificed 24 hours after administration of 2nd dose)
Histological examination
n=5 animals/sex/group (sacrificed 8 hours after administration of 1st dose)
n=5 animals/sex/group (sacrificed 24 hours after administration of 4th dose)

RESULTS

Brain of animal treated either with REL-1017 or with racemic methadone did not show the microscopic features of Olney's lesions

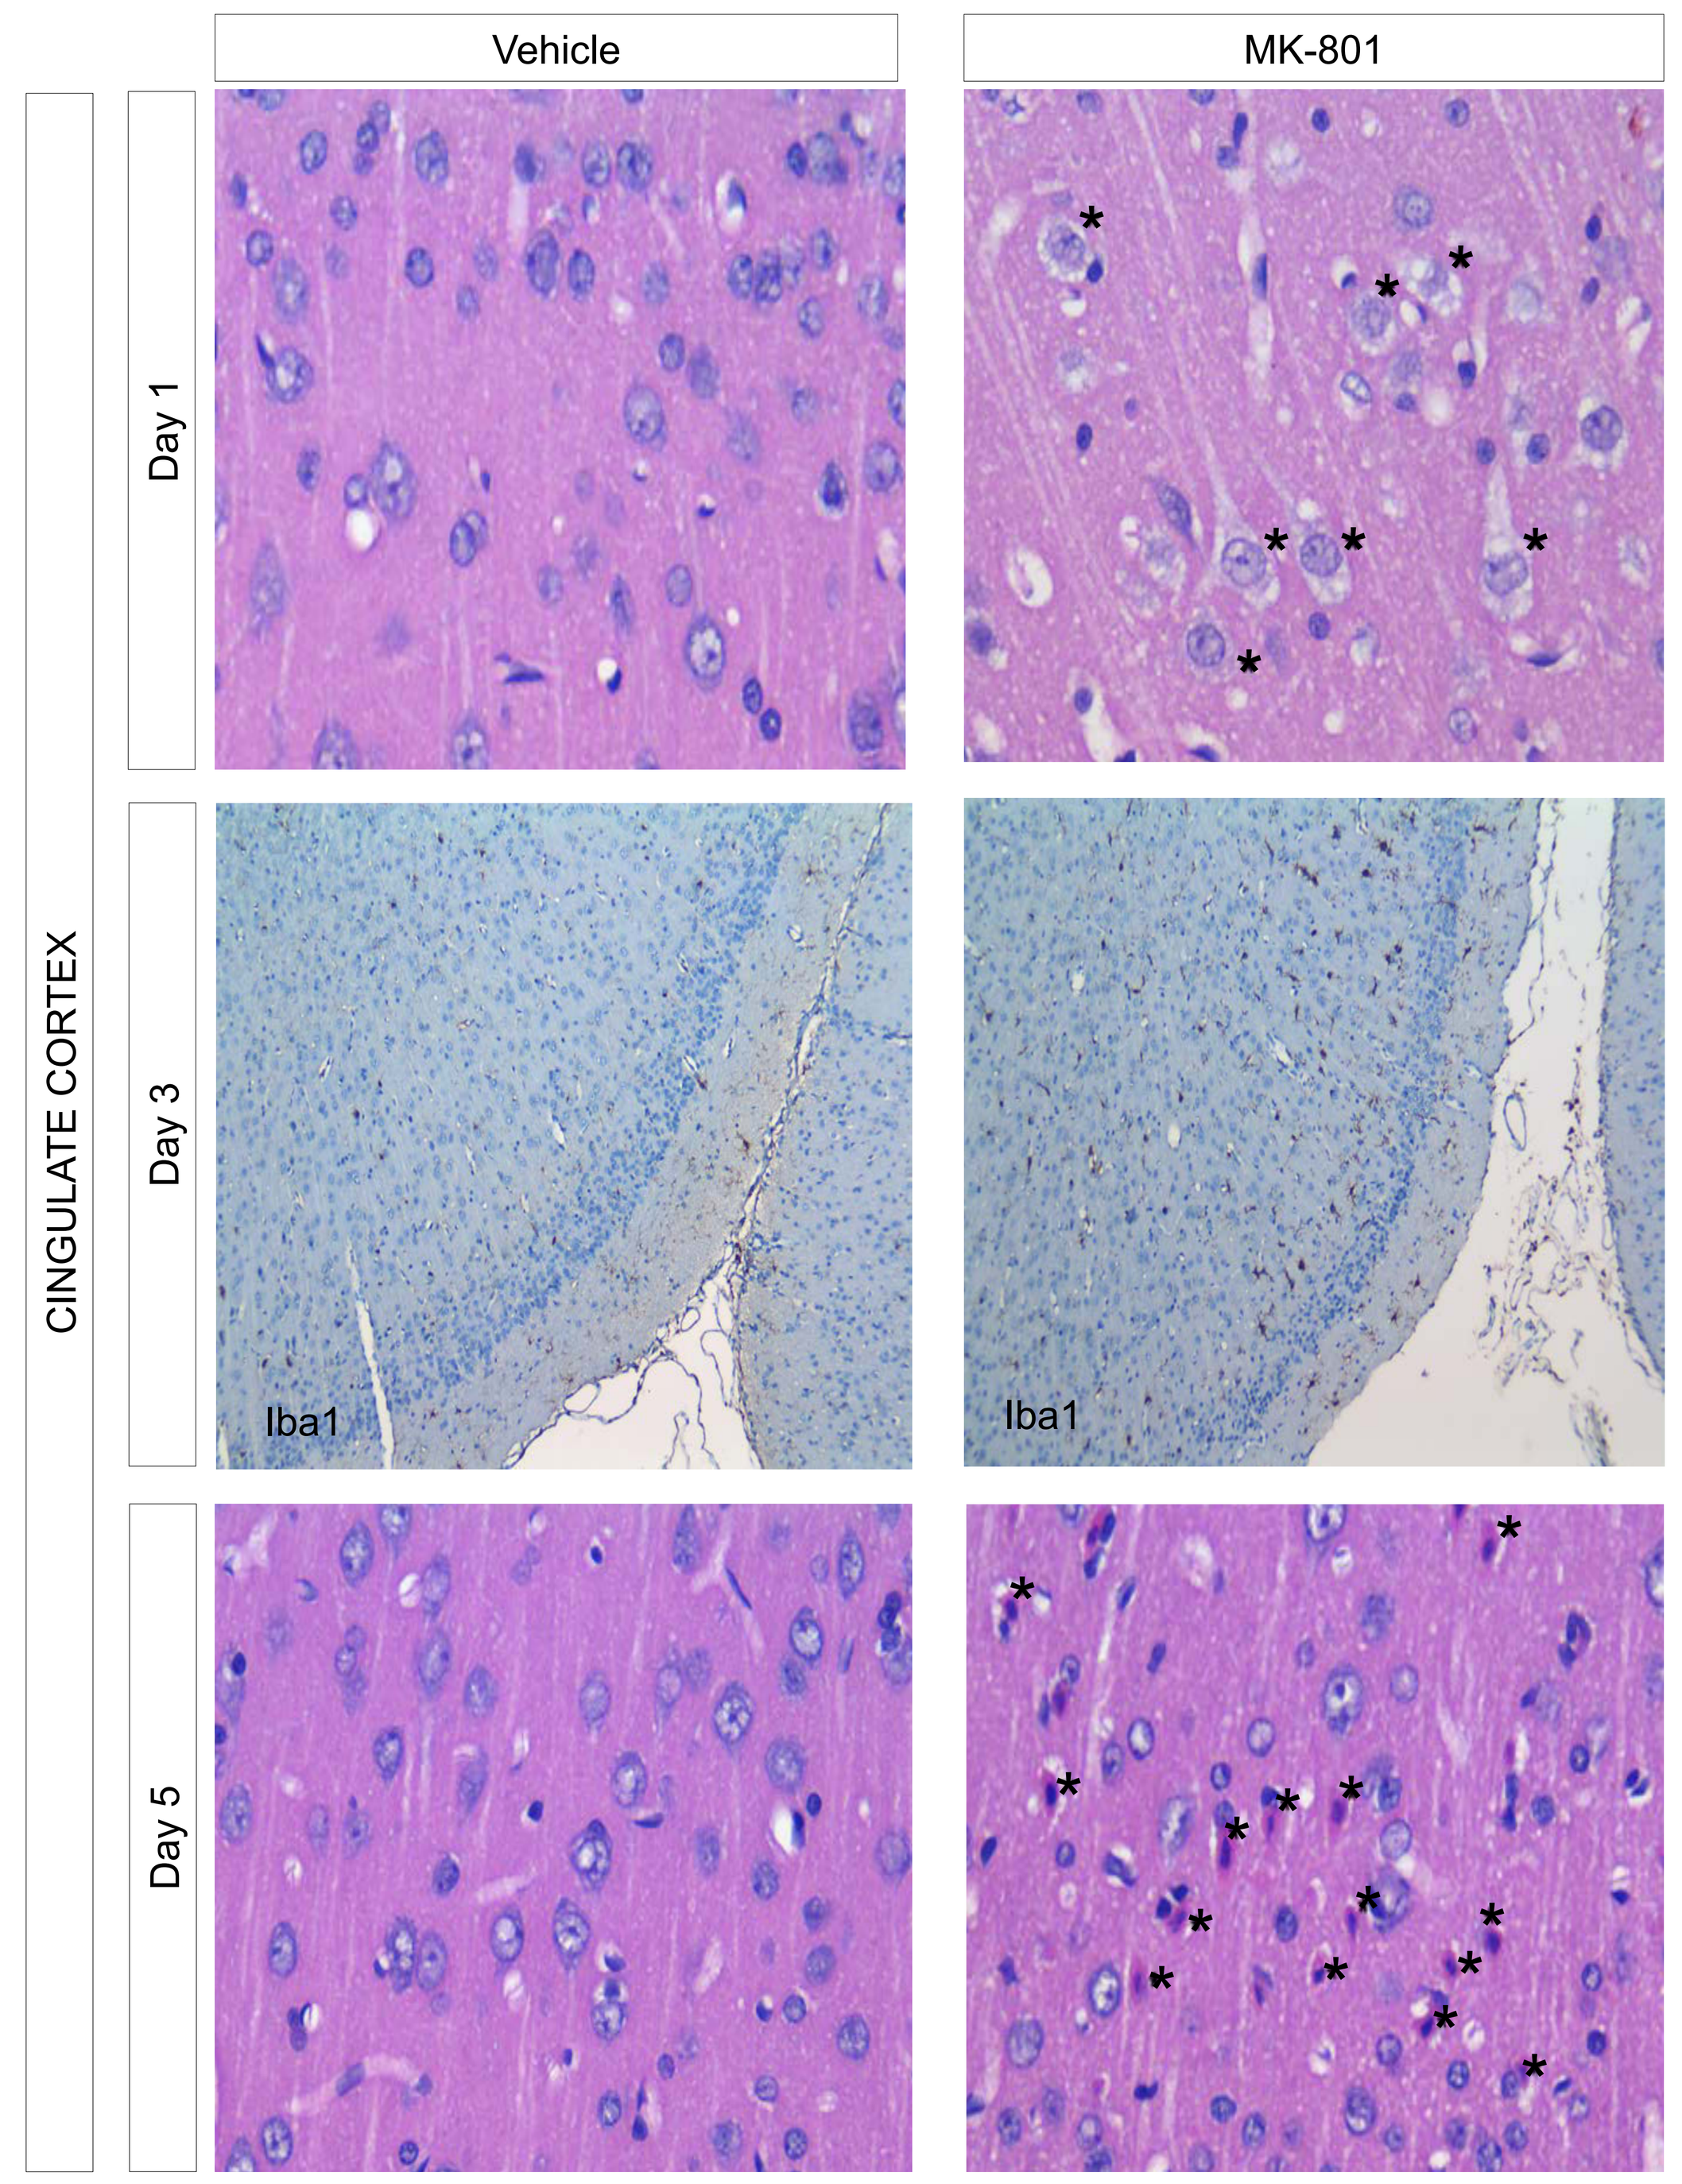
Vehicle
MK-801
CINGULATE CORTEX
Day 1
Day 3
Day 5
Iba1
Iba1

Neuronal vacuolation indicated by *

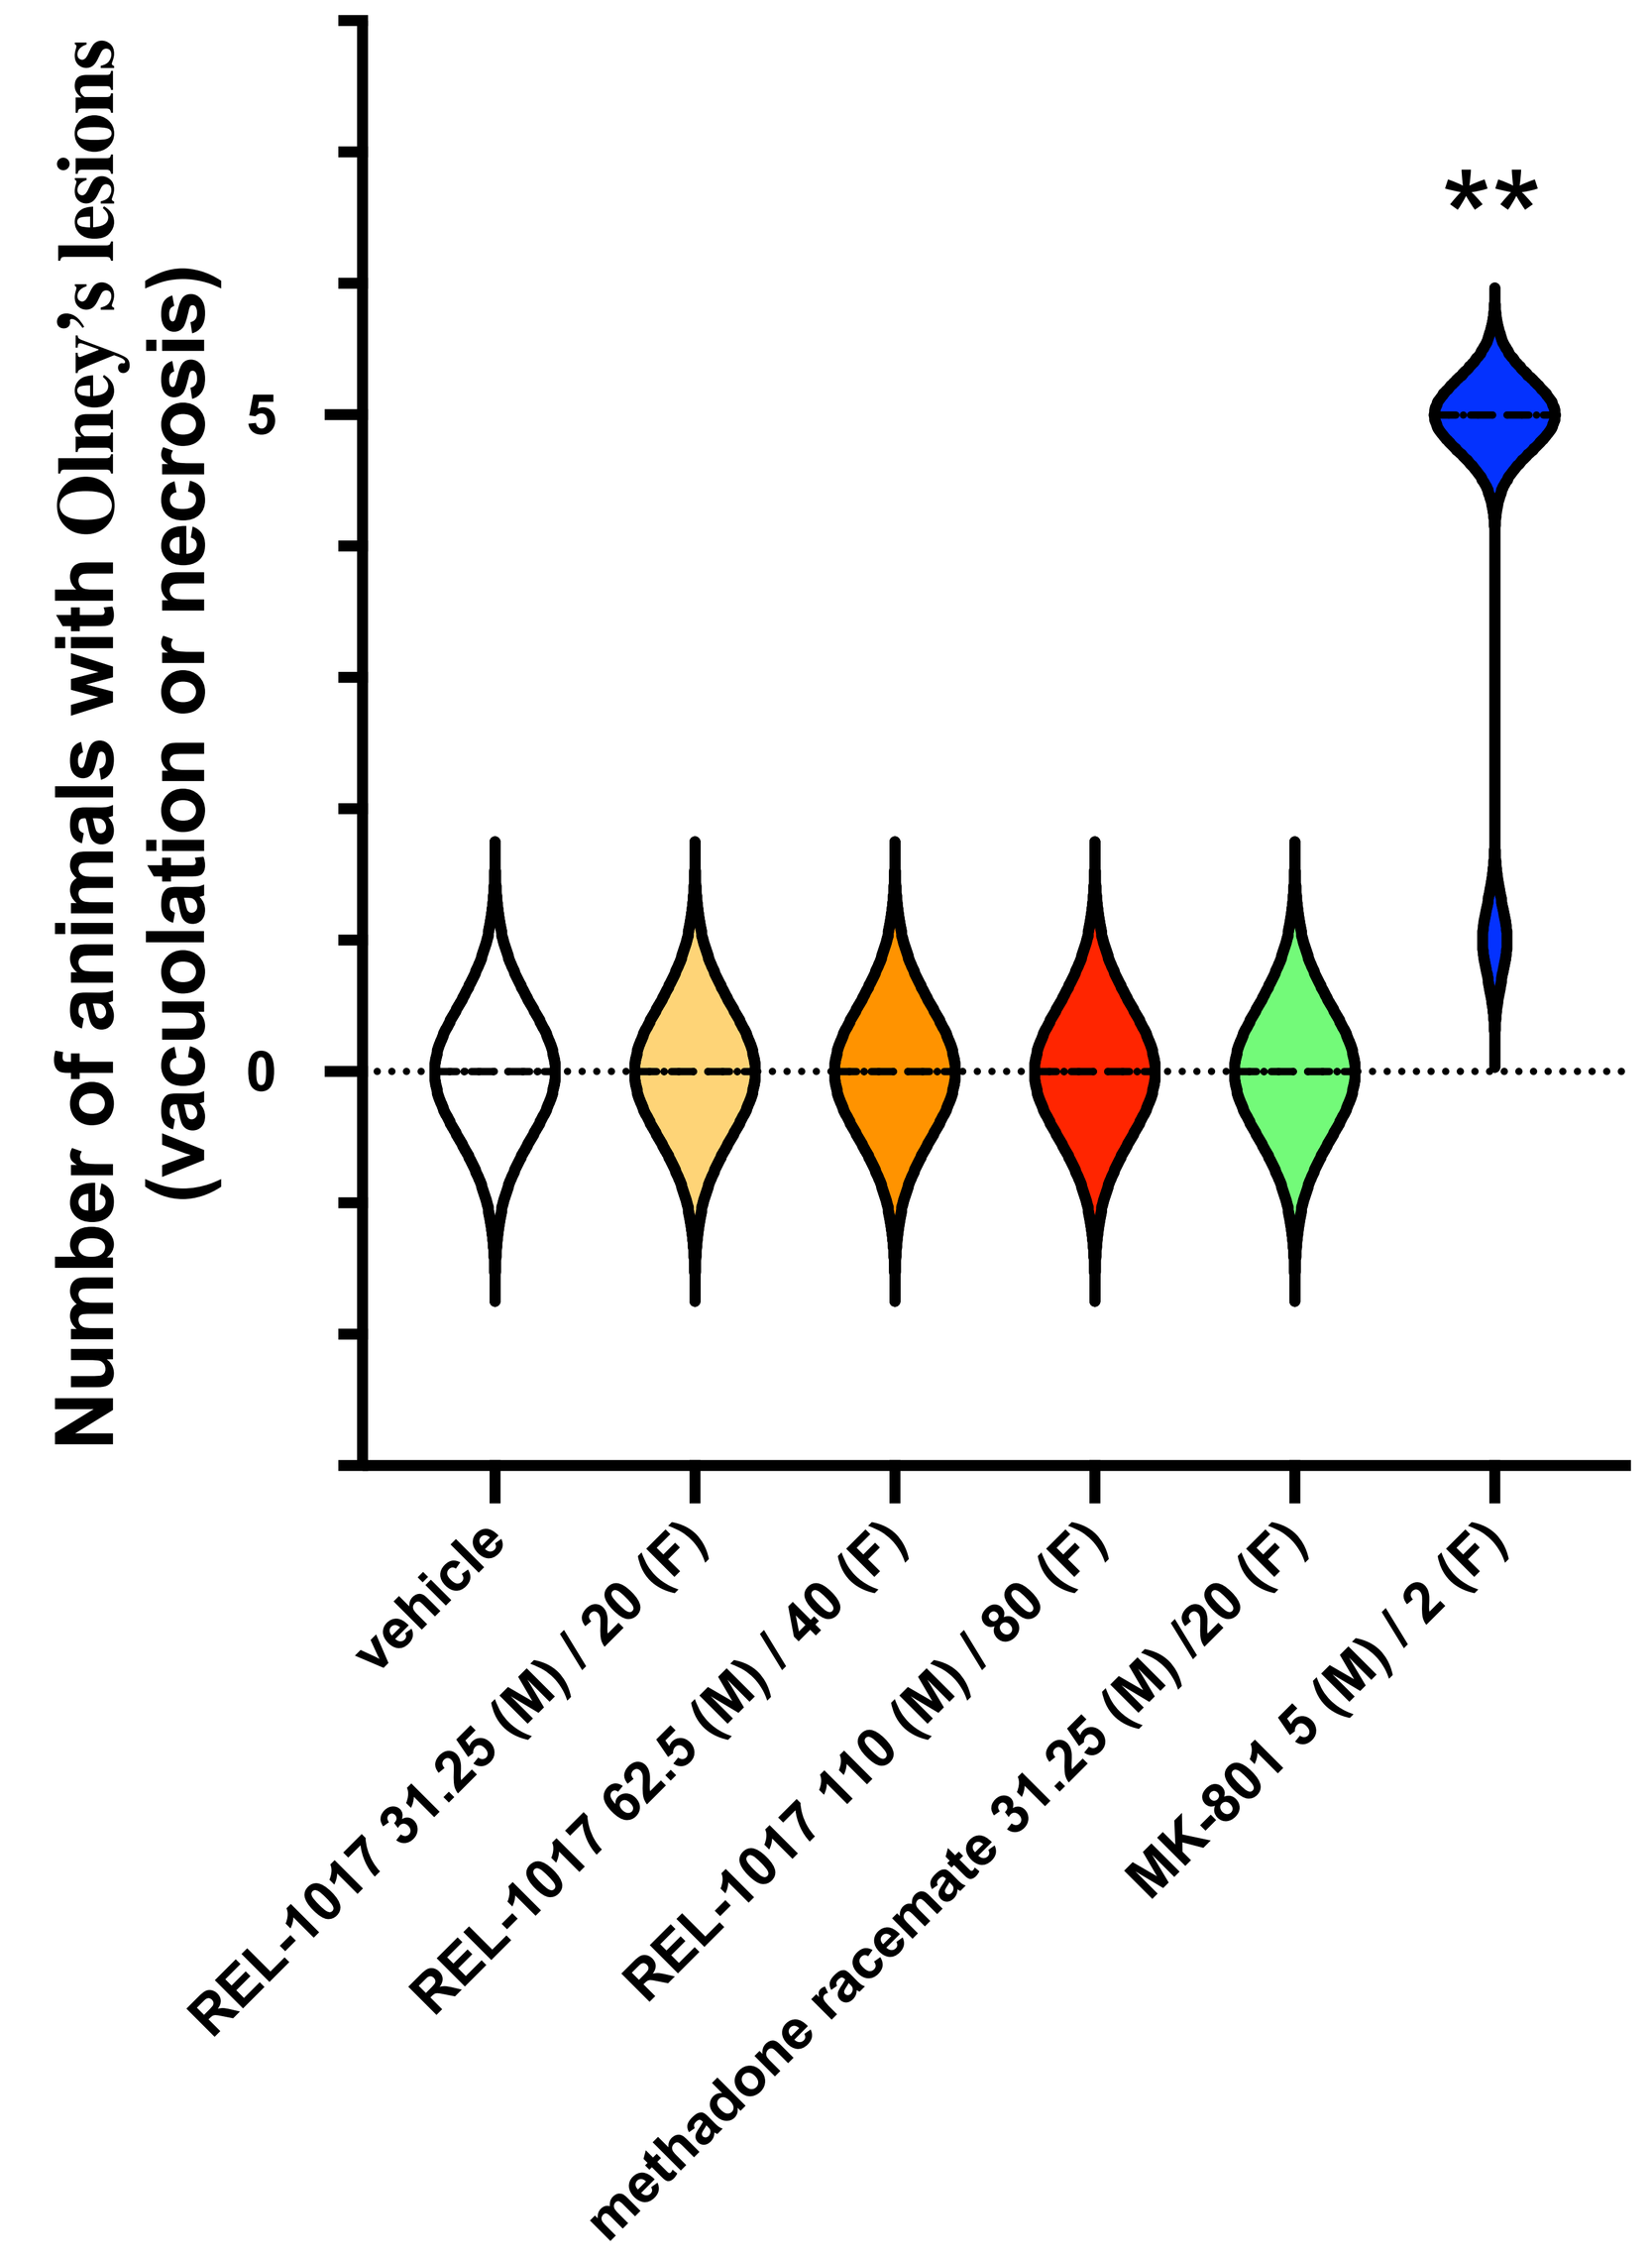
Number of animals with Olney's lesions (vacuolation or necrosis)
5
0
**
vehicle
REL-1017 31.25 (M) / 20 (F)
REL-1017 62.5 (M) / 40 (F)
REL-1017 110 (M) / 80 (F)
methadone racemate 31.25 (M) /20 (F)
MK-801 5 (M) / 2 (F)

The brain of animals treated with REL-1017 or with racemic methadone did not show the microscopic features of Olney's lesions. In contrast, the animals treated with MK-801 (positive control) showed, as expected, neuronal vacuolation and necrosis (hallmarks of Olney's lesions) (Fix, 1996)[3]. In particular, daily administration via oral gavage of REL-1017 for up to 4 days at all tested doses to both male and female SD rats did not produce Olney's lesions. Compared to REL-1017 at any dose and vehicle, the effects of MK-801 on cortical neurons were statistically significant ($p<0.01$). Furthermore, the oral administration of methadone racemate to both male and female SD rats did not produce Olney's lesions.

OBJECTIVES

To determine if the novel NMDAR antagonist REL-1017 administered once daily at three different doses for 4 days would cause cumulative neurotoxic effect and potential progression to irreversible damage (Olney's lesions) in the rat brain.

CONCLUSIONS

In this experiment, the administration of the novel NMDAR channel blocker REL-1017 (esmethadone) did not produce initial or cumulative neurotoxic effects or other evidence of damage to cortical neurons in normal rats.

REFERENCES

1. Olney JW et al., (1991) *Science,* 254 (5037)
2. Fogaça MV, et al., (2019) *Neuropsychopharmacology*, 44(13):2230-2238.
3. Fix, A. S., et al., (1996). *Toxicol Pathol, 24*(3), 291-304

DISCLOSURES

This research was sponsored by Relmada Therapeutics, Inc. Drs. Manfredi and Pappagallo are paid consultants of Relmada Therapeutics, Inc. Dr. Manfredi is an inventor on esmethadone patents and other patents and patent applications.

Relmada THERAPEUTICS

Corresponding Author: Paolo L. Manfredi, M.D. Email: pmanfredi@relmada.com