FRIDAY – 181

The Sub-MIC Effect of Lysin CF-301 on *Staphylococcus aureus (S. aureus)*

Jun Oh and **Raymond Schuch**

ContraFect Corporation, Yonkers, NY, USA

Contact Info:
R. Schuch, Ph.D.
ContraFect Corp.
914-207-2321
rschuch@contrafect.com

Introduction

CF-301 is a recombinantly-produced bacteriophage-derived lysin (cell wall hydrolase) and is the first agent of this class to enter clinical development for the treatment of bacteremia including endocarditis due to *S. aureus*.

To understand the activity of CF-301 at concentrations below the MIC, a series of *in vitro* pharmacodynamic parameters were previously examined in human serum. These studies demonstrated a strong post-antibiotic effect (PAE), post-antibiotic sub-MIC effect (PA-SME), and sub-MIC effect (SME) on a panel of 14 staphylococcal strains tested. We now report a more detailed study of the CF-301 SME on envelope ultrastructure, membrane permeability, membrane potential and describe how the SME impacts antibiotic susceptibility, biofilm formation, agglutination, *in vitro* growth and proliferation in a murine infection model.

Methods

The *in vitro* SME was induced over a range of CF-301 levels (1) using *S. aureus* strain MW2. The LIVE/DEAD BacLight Kit was used to determine variations in cell membrane permeability and the fluorescent reporter $DiSC_3$-5 was used to determine dissipation of membrane potential as described (2). A staphylococcal agglutination assay was performed using fibrinogen (3). Ultrastructural changes were examined by electron microscopy. The *in vivo* SME of CF-301 was determined in the neutropenic mouse thigh infection model (4). The SME on antibiotic susceptibility and biofilm formation was performed as indicated.

Study Outline

1. **Determine SME on features of the cell envelope:**
 a. Cell wall ultrastructure
 b. Membrane integrity
 c. Membrane potential
2. **Determine SME on critical staphylococcal phenotypes:**
 a. Antibiotic susceptibility – Daptomycin (DAP)
 b. Biofilm formation
 c. Agglutination
 d. Growth (*in vitro* and *in vivo*)

References

1. Pankuch and Applebaum, 2006. Antimicrob. Agents Chemother. 50:3956-3958.
2. Berti, et al., 2012. Antimicrob. Agents Chemother. 56:5046-5053.
3. Walker, et al., 2013. PLoS Pathog. 9:1-17
4. Vogelman, et al., 1988. J. Infect. Dis. 157:287-298

1. SME on the cell envelope

We hypothesize that when concentrations fall below the MIC, the cell wall hydrolytic activity of CF-301 still causes significant (albeit non-lethal) damage to the cell envelope. The previously reported mean SME in human serum of up to 7.8 hours, must represent the time needed to repair damage and resume growth. Here, we examine the nature of this damage over a sub-MIC range.

1a. Cell envelope ultrastructure

Mid-log phase cultures of *S. aureus* strain MW2 exposed to CF-301 (at indicated xMIC* levels) for 1 hour in human serum

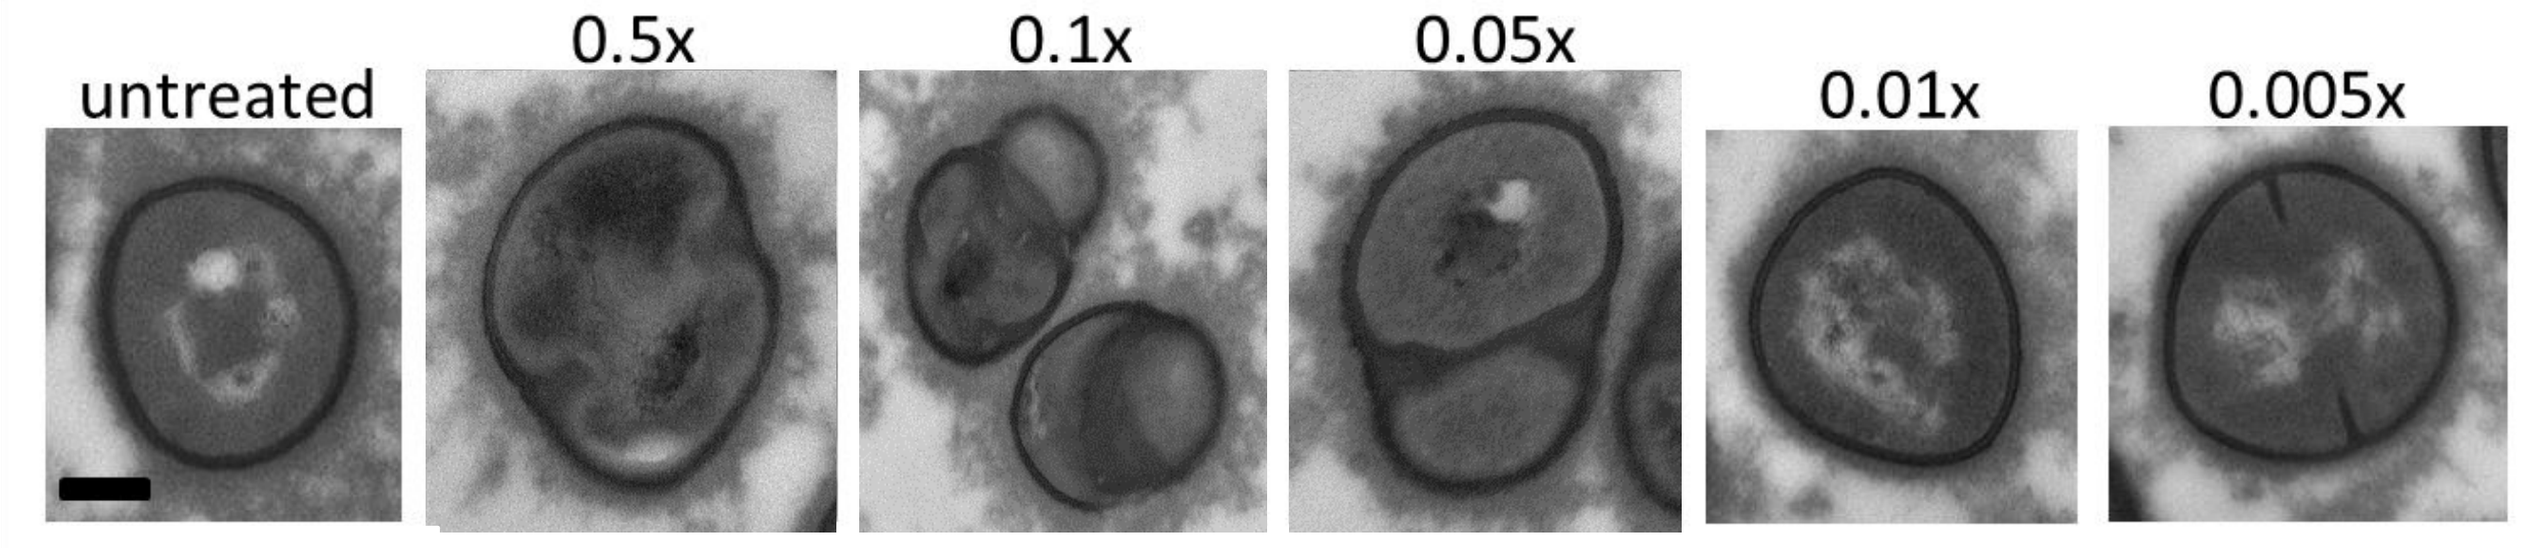
untreated
0.5x
0.1x
0.05x
0.01x
0.005x
13 kX,
Scale = 200 nm
Aberrant septation

*1x MIC = 1 μg/mL

1b. Membrane integrity

Mid-log phase *S. aureus* strain MW2 was exposed to CF-301, DAP, or CF-301+DAP (indicated xMIC values) for 2 hours at 24°C prior to analysis. **(A)** Staining with LIVE/DEAD (Syto9/PI) BacLight Kit. A lower value indicates more extensive membrane damage. **(B)** Bacterial viability. In each case, means of 3 experiments are shown. 1X MIC = 1 μg/ml and 8 μg/ml for CF-301 and DAP, respectively.

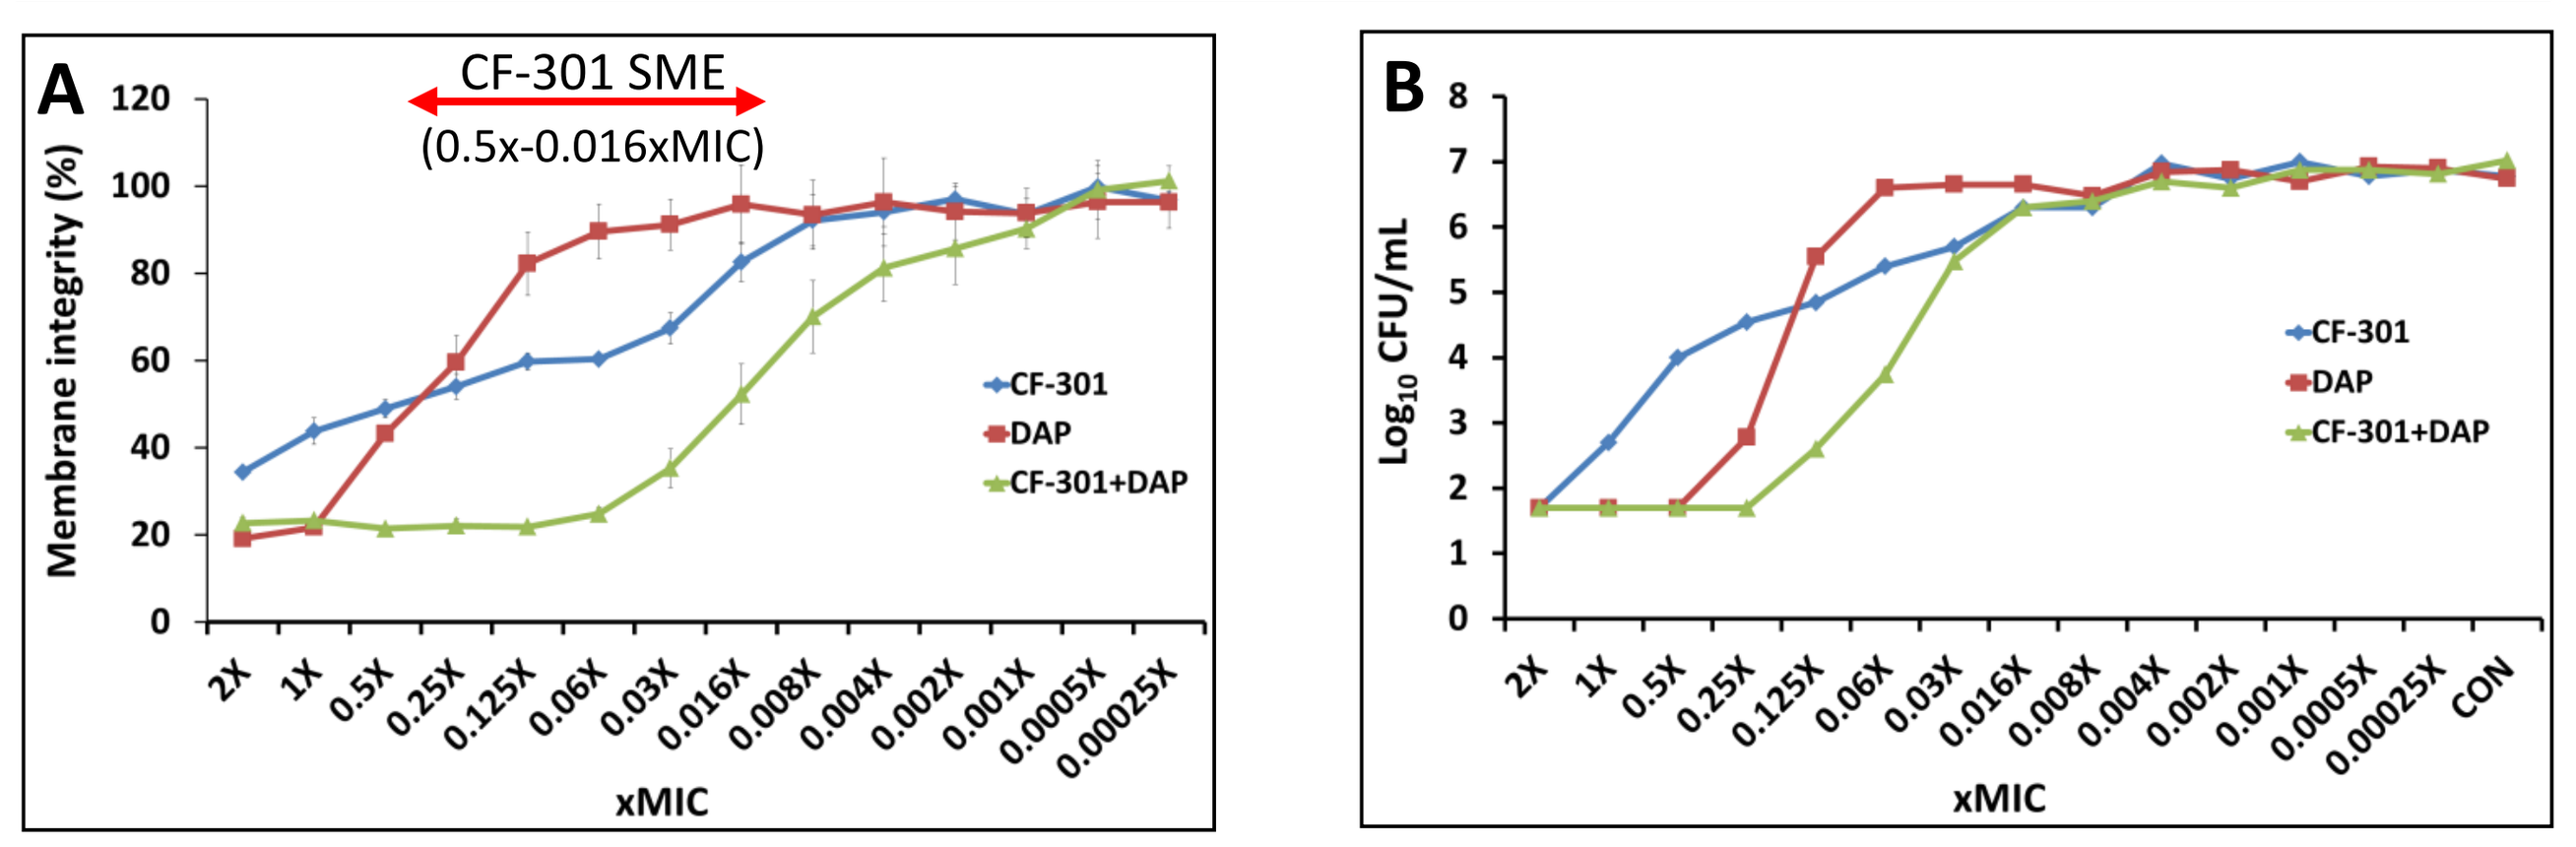
A
CF-301 SME
(0.5x-0.016xMIC)
Membrane integrity (%)
xMIC
CF-301
DAP
CF-301+DAP
B
Log10 CFU/mL
xMIC
CF-301
DAP
CF-301+DAP

- The SME extends to the following concentrations: CF-301 = 0.016x MIC; DAP = 0.12x MIC; and CF-301+DAP = 0.004x
- SME extends below concentrations affecting overall viability

1c. Membrane potential (single agent)

Mid-log phase *S. aureus* MW2 was stained with $DiSC_3$-5 and exposed to CF-301 **(A)** and DAP **(B)** for a 2 hour kinetic analysis of $DISC_3$-5 release from depolarized membranes. An increase in signal indicates depolarization.

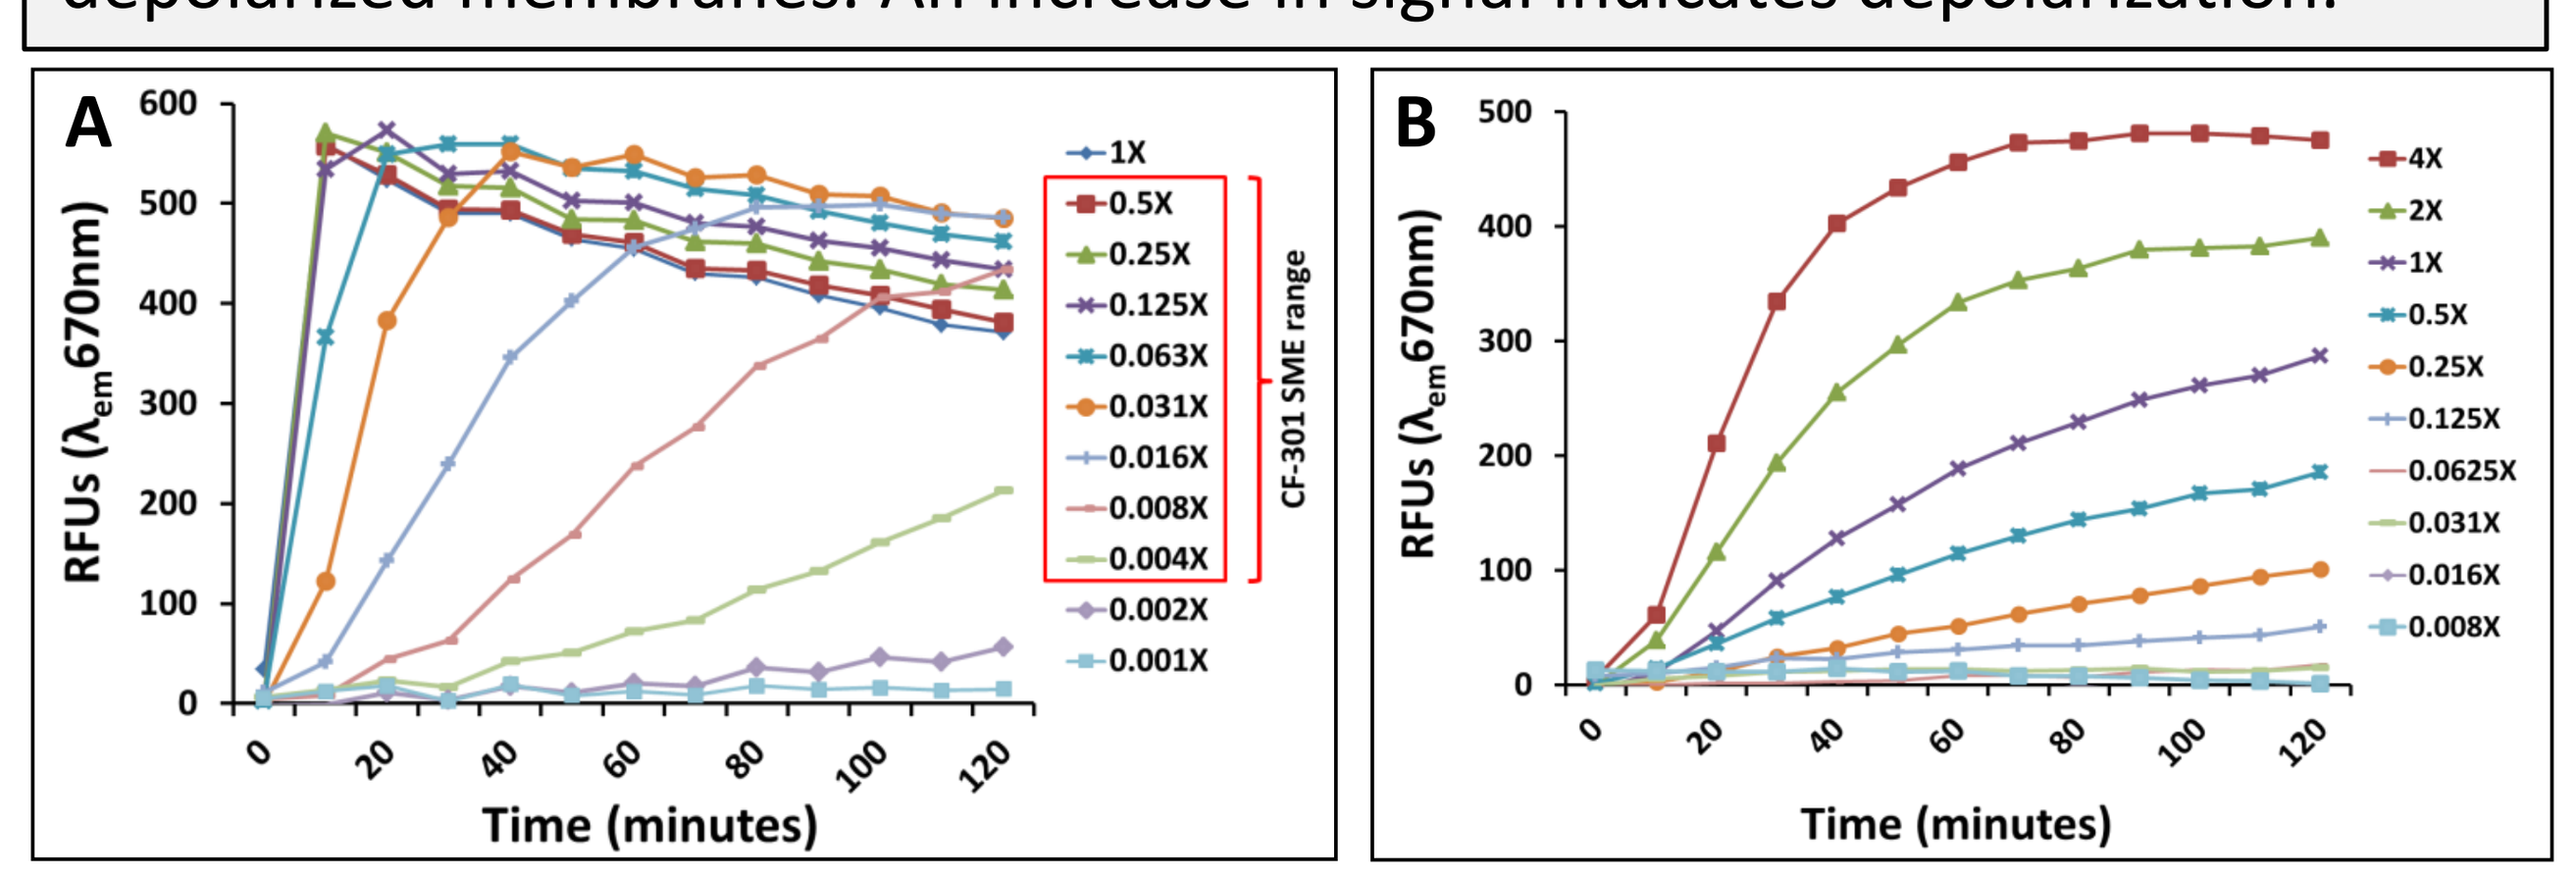
A
RFUs (λem670nm)
Time (minutes)
1X
0.5X
0.25X
0.125X
0.063X
0.031X
0.016X
0.008X
0.004X
0.002X
0.001X
CF-301 SME range
B
RFUs (λem670nm)
Time (minutes)
4X
2X
1X
0.5X
0.25X
0.125X
0.0625X
0.031X
0.016X
0.008X

- CF-301 exerts a potent effect on membrane stability over a broad sub-MIC range (0.5x-0.004x)
- The effect is similar to that of DAP at very low concentrations

1c. Membrane potential (dual agent)

Mid-log phase *S. aureus* MW2 was stained with $DiSC_3$-5 and exposed to CF-301 in combinations with DAP. **(A)** Single agent and dual agent analysis. **(B)** Fixed [CF-301], with a range of [DAP]. **(C)** Fixed [DAP], with a range of [CF-301].

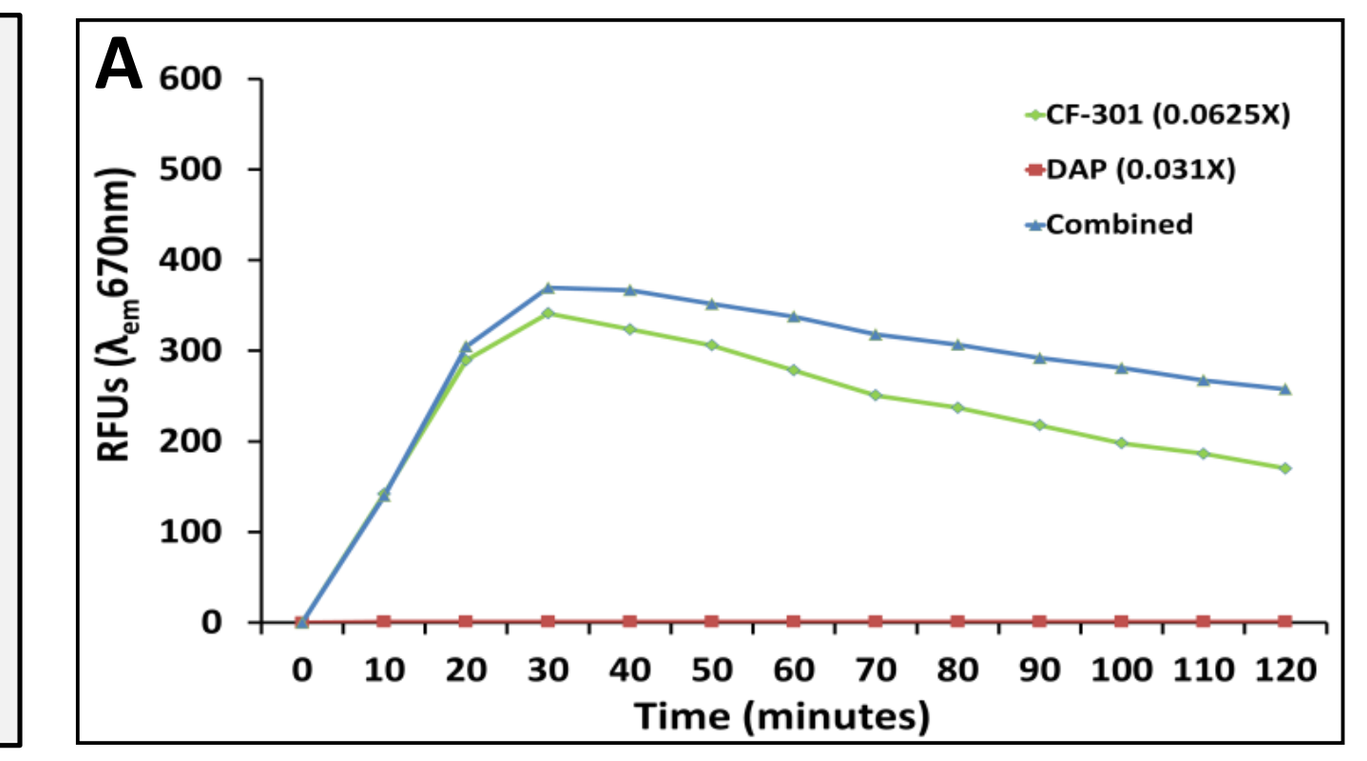
A
RFUs (λem670nm)
Time (minutes)
CF-301 (0.0625X)
DAP (0.031X)
Combined

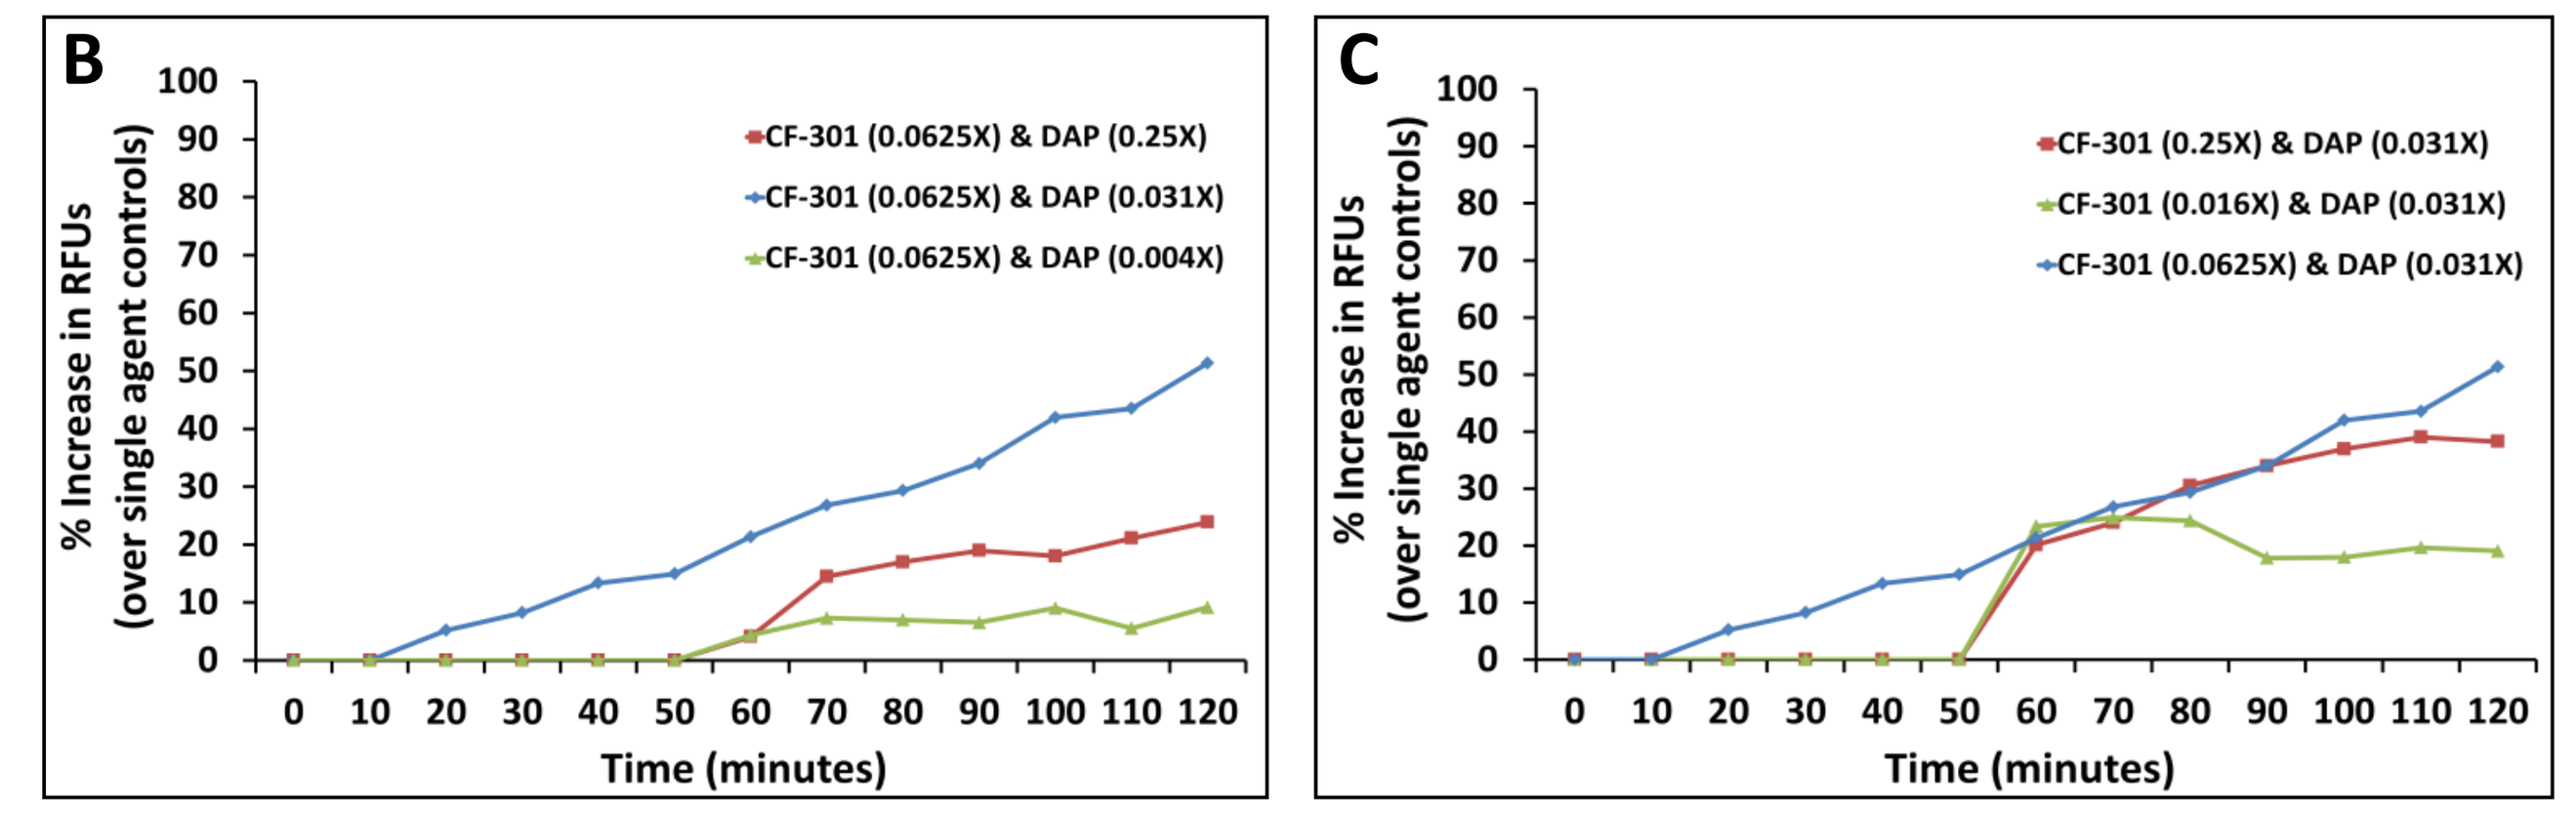
B
% Increase in RFUs
(over single agent controls)
Time (minutes)
CF-301 (0.0625X) & DAP (0.25X)
CF-301 (0.0625X) & DAP (0.031X)
CF-301 (0.0625X) & DAP (0.004X)
C
% Increase in RFUs
(over single agent controls)
Time (minutes)
CF-301 (0.25X) & DAP (0.031X)
CF-301 (0.016X) & DAP (0.031X)
CF-301 (0.0625X) & DAP (0.031X)

- Combinations of CF-301 and DAP at low sub-MIC levels can enhance the disruption of membrane potential

2. SME on staphylococcal phenotypes

The SME of CF-301 on the cell envelope is expected to have a range of different downstream effects on antibiotic susceptibility, biofilm formation, agglutination, and growth *in vitro* and *in vivo*.

2a. Antibiotic susceptibility

Mid-log phase cultures of *S. aureus* strain MW2 exposed to CF-301 or DAP (xMIC levels) for 1 hour in human serum, then diluted 1:1000 into the broth microdilution assay for the indicated agent.

SME induced with CF-301

[CF-301] (xMIC)	Fold reduction (DAP MIC)
0.5	>8
0.25	>8
0.125	8
0.06	4
0.03	4
0.015	2
0.008	2

SME induced with DAP

SME induction (xMIC DAP)	Fold reduction (CF-301 MIC)
0.75	8
0.4	4
0.2	4
0.1	4
0.05	4
0.025	2
0.013	2

- The CF-301 SME resulted in enhanced susceptibility to DAP
- The DAP SME resulted in enhanced susceptibility to CF-301

2b. Biofilm formation

Mid-log phase *S. aureus* strain ATCC BAA-42 was exposed to CF-301 (xMIC levels) for 1 hour in human serum, and diluted 1:1000 into the biofilm assay. The PAE or SME is defined as the time required for biofilm biomass in each treatment to reach 75% of the biofilm biomass observed in the untreated control over a 36 hour time course.

PAE/SME induction (xMIC CF-301)	Mean effect (hours)	
4	20	PAE
2	8	PAE
1	8	PAE
0.5	7	SME
0.25	6	SME
0.1	3	SME
0.05	0	

- The CF-301 SME (and PAE) acted to delay the formation of *in vitro* biofilms

2c. Agglutination

Mid-log phase *S. aureus* MW2 was exposed to CF-301 (xMIC shown) for 2 hours and then suspended in PBS with fibrinogen. A reduction in culture optical density is monitored as agglutination causes the biomass to settle.

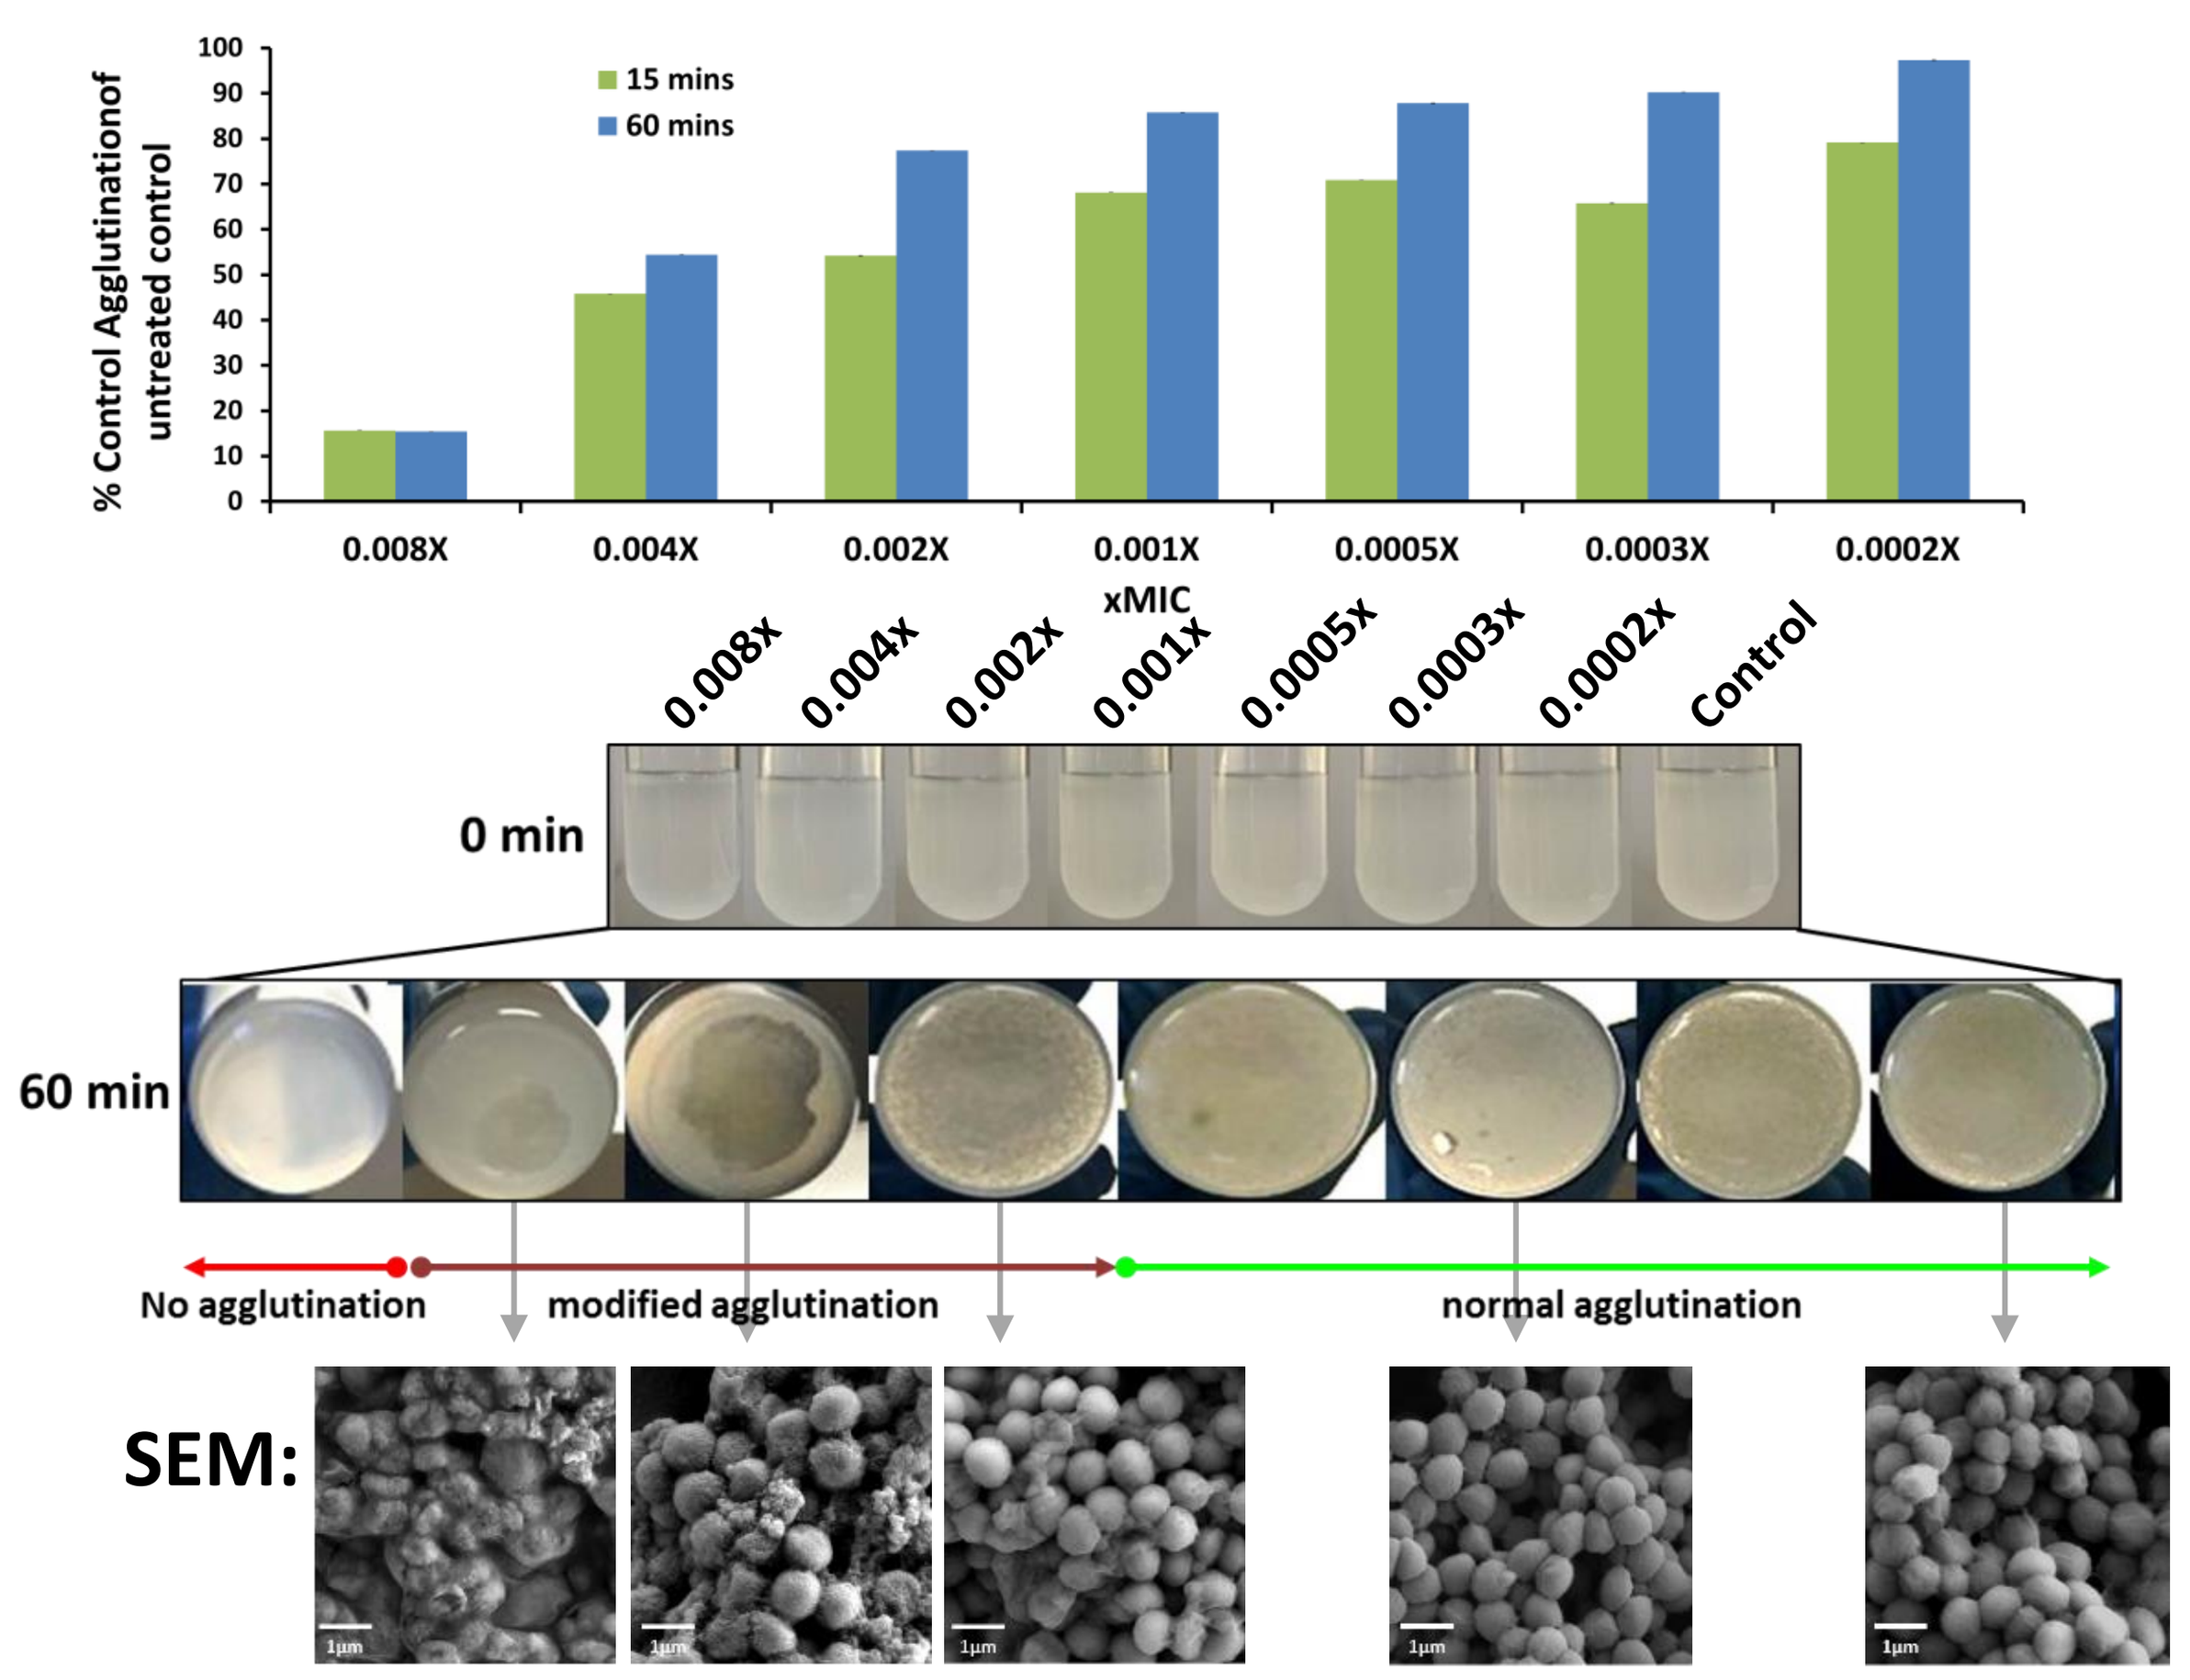
% Control Agglutinationof untreated control
15 mins
60 mins
0.008X
0.004X
0.002X
0.001X
0.0005X
0.0003X
0.0002X
xMIC
0.008x
0.004x
0.002x
0.001x
0.0005x
0.0003x
0.0002x
Control
0 min
60 min
No agglutination
modified agglutination
normal agglutination
SEM:

- A potent SME on agglutination was observed

2d. Growth

In vitro Analysis: *S. aureus* strain MW2 exposed to CF-301 for 1 hour in human serum, then diluted 1:100 into 0.5x LB and followed growth over for 24 hours

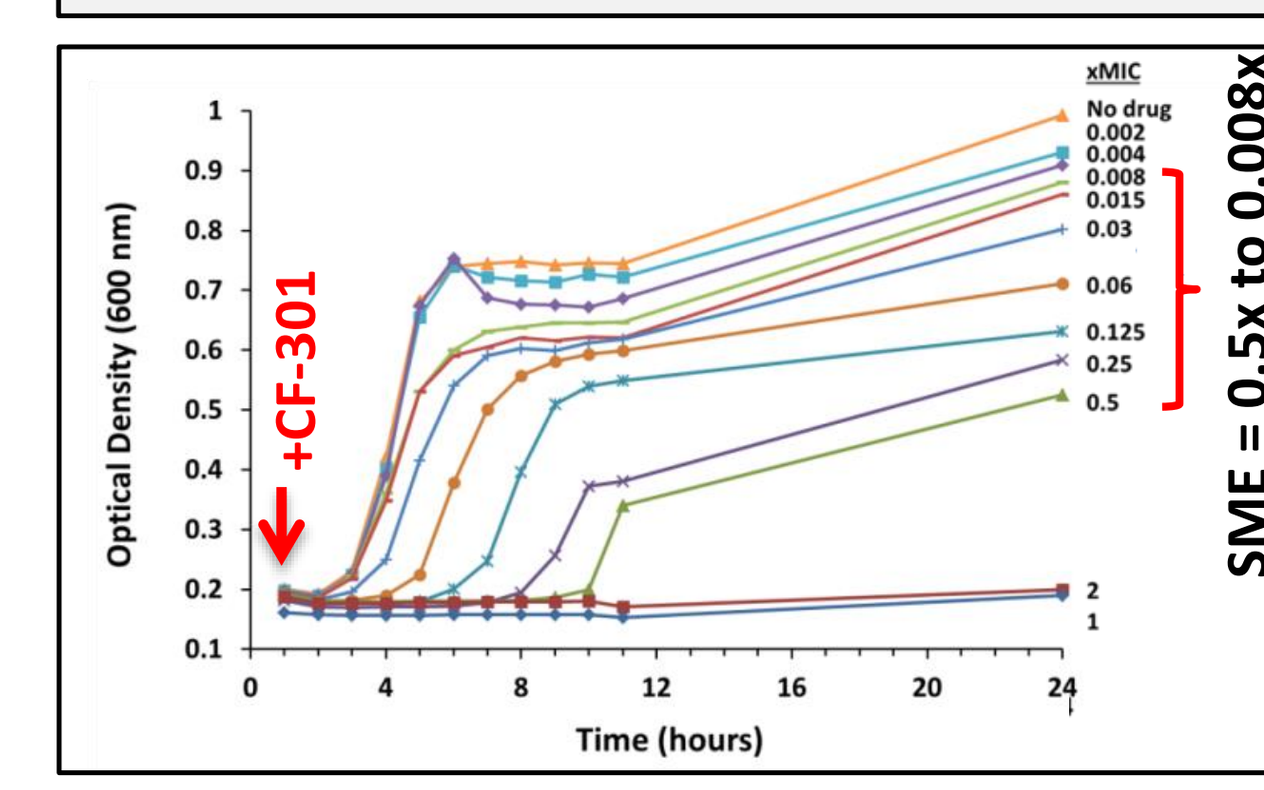
Optical Density (600 nm)
Time (hours)
+CF-301
xMIC
No drug
0.002
0.004
0.008
0.015
0.03
0.06
0.125
0.25
0.5
2
1
SME = 0.5x to 0.008x

- Exposure to CF-301 over a broad sub-MIC range had an inhibitory effect on growth, extending lag phase and shortening exponential phase

In vivo Analysis: Growth was examined in the neutropenic mouse thigh model using two *S. aureus* strains. SME is the time for a 1-log increase in CFUs in CF-301-treated mice minus the corresponding time in control mice.

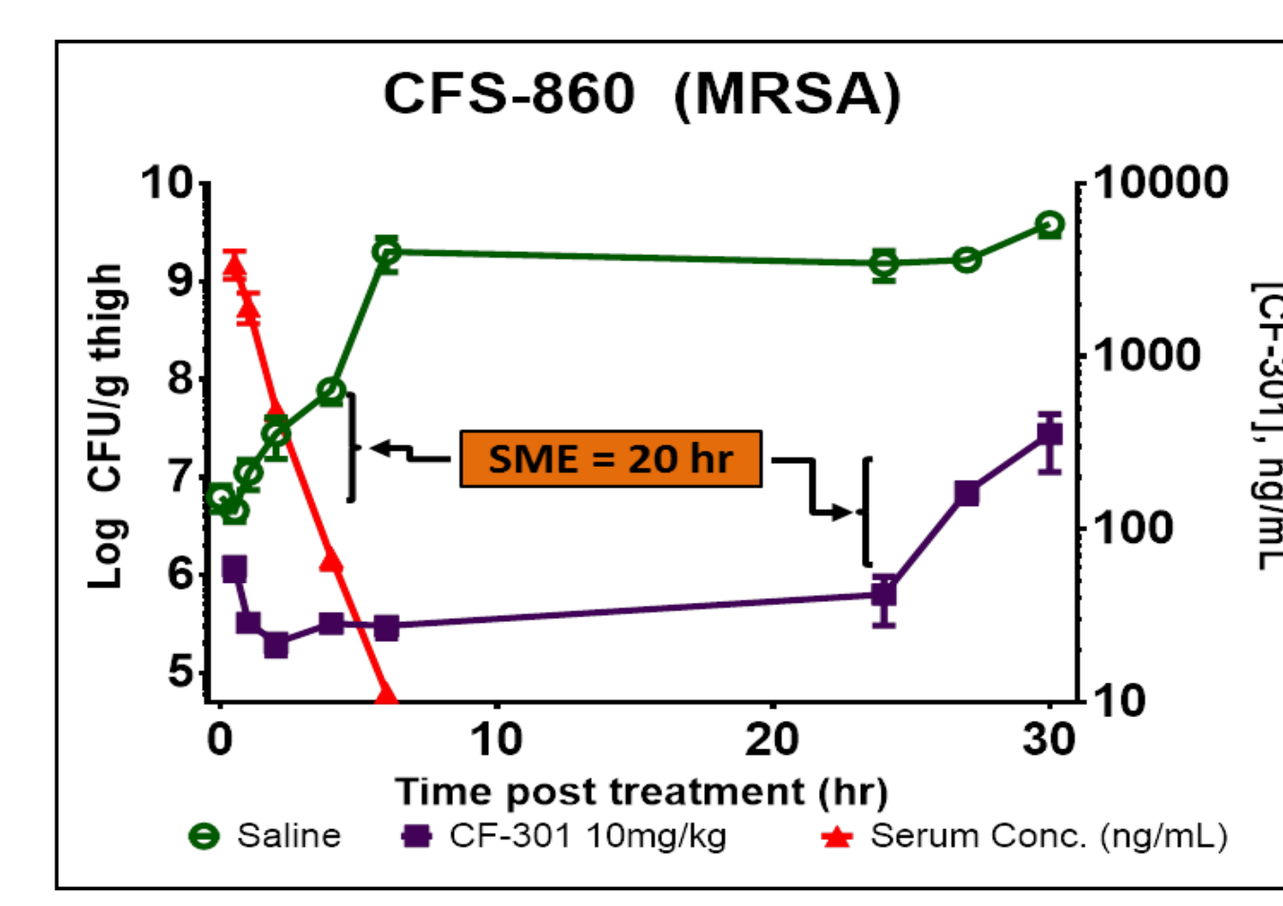
CFS-860 (MRSA)
Log CFU/g thigh
[CF-301], ng/mL
SME = 20 hr
Time post treatment (hr)
Saline
CF-301 10mg/kg
Serum Conc. (ng/mL)

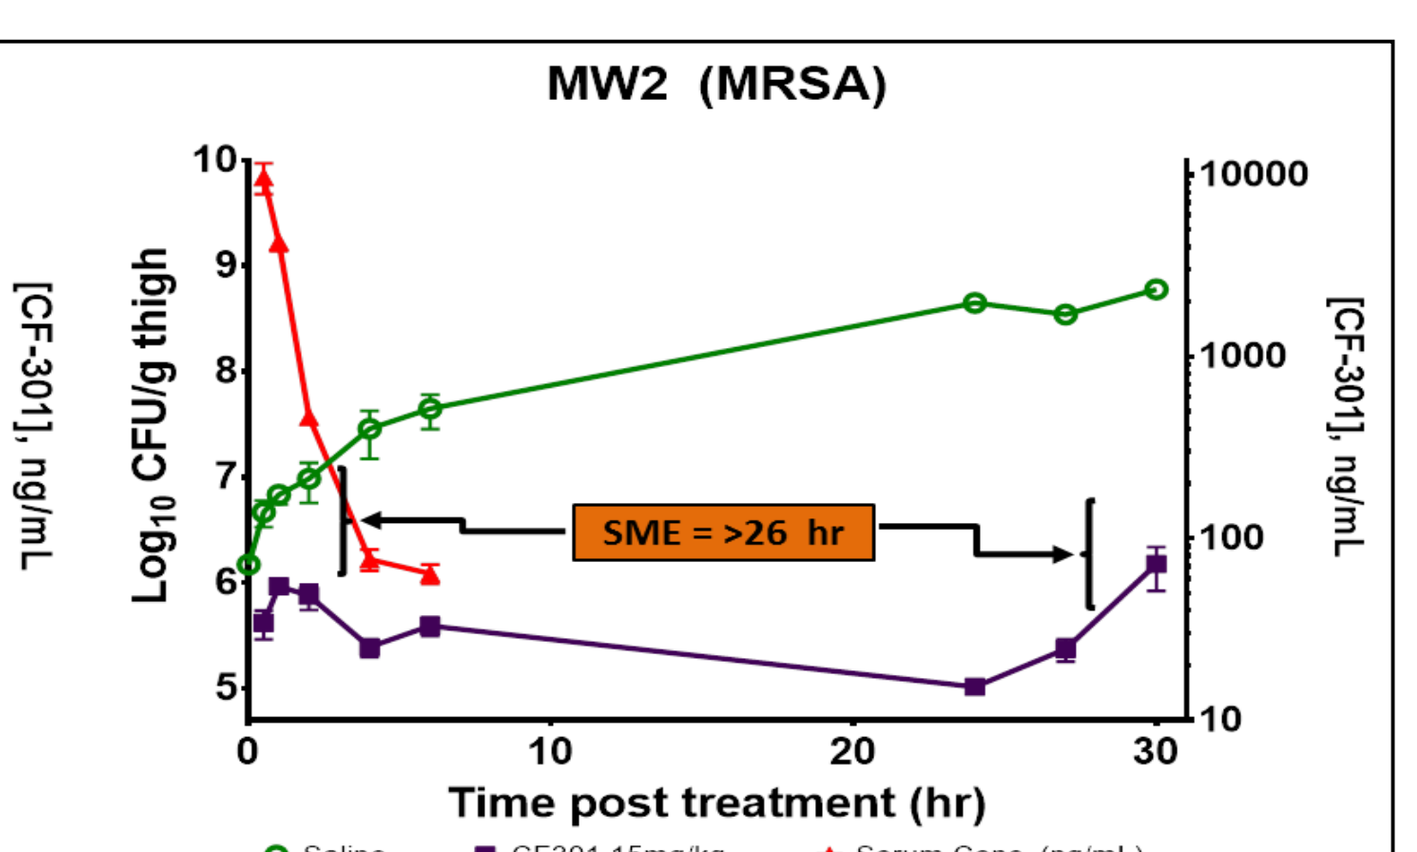
MW2 (MRSA)
Log10 CFU/g thigh
[CF-301], ng/mL
SME = >26 hr
Time post treatment (hr)
Saline
CF301 15mg/kg
Serum Conc. (ng/mL)

- A potent *in vivo* SME was observed in the context of infect mice

Conclusions

- Exposure to CF-301 levels as low as 0.004x MIC increased antibiotic susceptibility, decreased biofilm formation, inhibited a virulence phenotype, and reduced growth rates in vitro and in infected mice
- As CF-301 levels fall below the MIC, sustained inhibition of antibiotic resistance and biofilm formation is anticipated to improve the efficacy of antistaphylococcal antibiotics used in combination with CF-301.