IBREXAFUNGERP (SCY-078): A FIRST-IN-CLASS, ORALLY-BIOAVAILABLE, GLUCAN SYNTHASE INHIBITOR HAS FUNGICIDAL ACTIVITY AGAINST *C. AURIS,* AN EMERGING MULTIDRUG-RESISTANT PATHOGEN

Stephen Barat[1], Katyna Borroto-Esoda[1], David Angulo[1]
SCYNEXIS, Inc., USA

BACKGROUND

Ibrexafungerp (formerly SCY-078) is a novel, oral and intravenous, triterpenoid class glucan synthase inhibitor with broad-spectrum activity against *Candida, Aspergillus and Pneumocystis* spp. currently in clinical development for use in the treatment of various fungal infections. *C. auris* is an emerging, multi-drug resistant pathogen, with some isolates resistant to azoles, polyenes and echinocandins. The purpose of this effort was to evaluate the *in vitro* activity of Ibrexafungerp against *C. auris*.

METHODS

The *in vitro* activity of Ibrexafungerp was evaluated in two independent studies against >110 clinical *C. auris* isolates obtained from various countries including India, Pakistan, Columbia, South Africa and the United States. The studies included 7 isolates with elevated MICs to the echinocandins. *In vitro* susceptibility was determined by broth micro-dilution using CLSI methods (M27-A3, 50% inhibition at 24 hours); comparator compounds included micafungin (MFG), caspofungin (CAS) and anidulafungin (AFG). .

To evaluate the activity of Ibrexafungerp against *C. auris* biofilms, discs with mature biofilms were transferred to wells containing different concentrations of SCY-078 (ranging from 0.5 to 4 μg/L). Following 48 h incubation, metabolic activities of biofilms were measured using the XTT reduction assay. Images and thickness of biofilms growing in the presence or absence of drug were captured using confocal scanning laser microscopy (CSLM)

The effect of Ibrexafungerp on *C. auris* morphology and ultrastructure was determined using scanning electron microscopy (SEM) and transmission electron microscopy (TEM). *C. auris* strains were exposed overnight to 1x MIC (0.5 mg/L) of Ibrexafungerp, cell suspensions where then fixed in 2% glutaraldehyde and incubated at 4 °C for 48 h. After fixation, samples were processed and dried. Processed samples were coated with palladium for 60s and viewed with a Nova NanoLab 200 FEG-SEM/FIB scanning electron microscope in high-vacuum mode at 2.00 kV.

RESULTS

Ibrexafungerp (IBX) demonstrated fungicidal activity *in vitro* against all clades of *C. auris* with no meaningful variation in MICs (0.5-1.0 ug/ml) across studies (Table 1), including those strains with elevated MICs to echinocandins (Table 2)

Table 1 - MIC (μg/mL) Distribution of Ibrexafungerp and comparators vs *C. auris* clinical isolates

CDC[a]		Center for Medical Mycology[b]				
N (100)	IBX	N (16)	IBX	CAS	MFG	AFG
Range	0.0625 - 2	Range	0.5 - 2	0.25 – 1	0.25 - 2	0.125 – 0.25
Mode	1	Mode	1	0.5	1	0.125
MIC_{50}	0.5	MIC_{50}	1	0.5	1	0.125
MIC_{90}	1	MIC_{90}	1	1	2	0.25

[a]Berkow EL et al., AAC 2017, [b]Larkin E et al., AAC 2017

Table 2 – Activity of Ibrexafungerp (SCY-078) against *C. auris* isolates with elevated MICs to the Echinocandins

	MIC (μg/mL) of drug[a]			
Isolate	IBX	Anidulafungin	Caspofungin	Micafungin
1	1	8	1	4
2	1	16	1	4
3	1	1	16	1
4	1	2	16	2
5	0.5	4	0.5	0.5
6	0.5	>16	>16	>8
7	1	4	>16	1

[a]Berkow EL et al., AAC 2017

RESULTS

Ibrexafungerp (SCY-078) demonstrated activity against *C. auris* biofilms, as evidenced by reductions in biofilm thickness and metabolic activity.

Control | 0.5 μg/ml | 2 μg/ml | 4 μg/ml

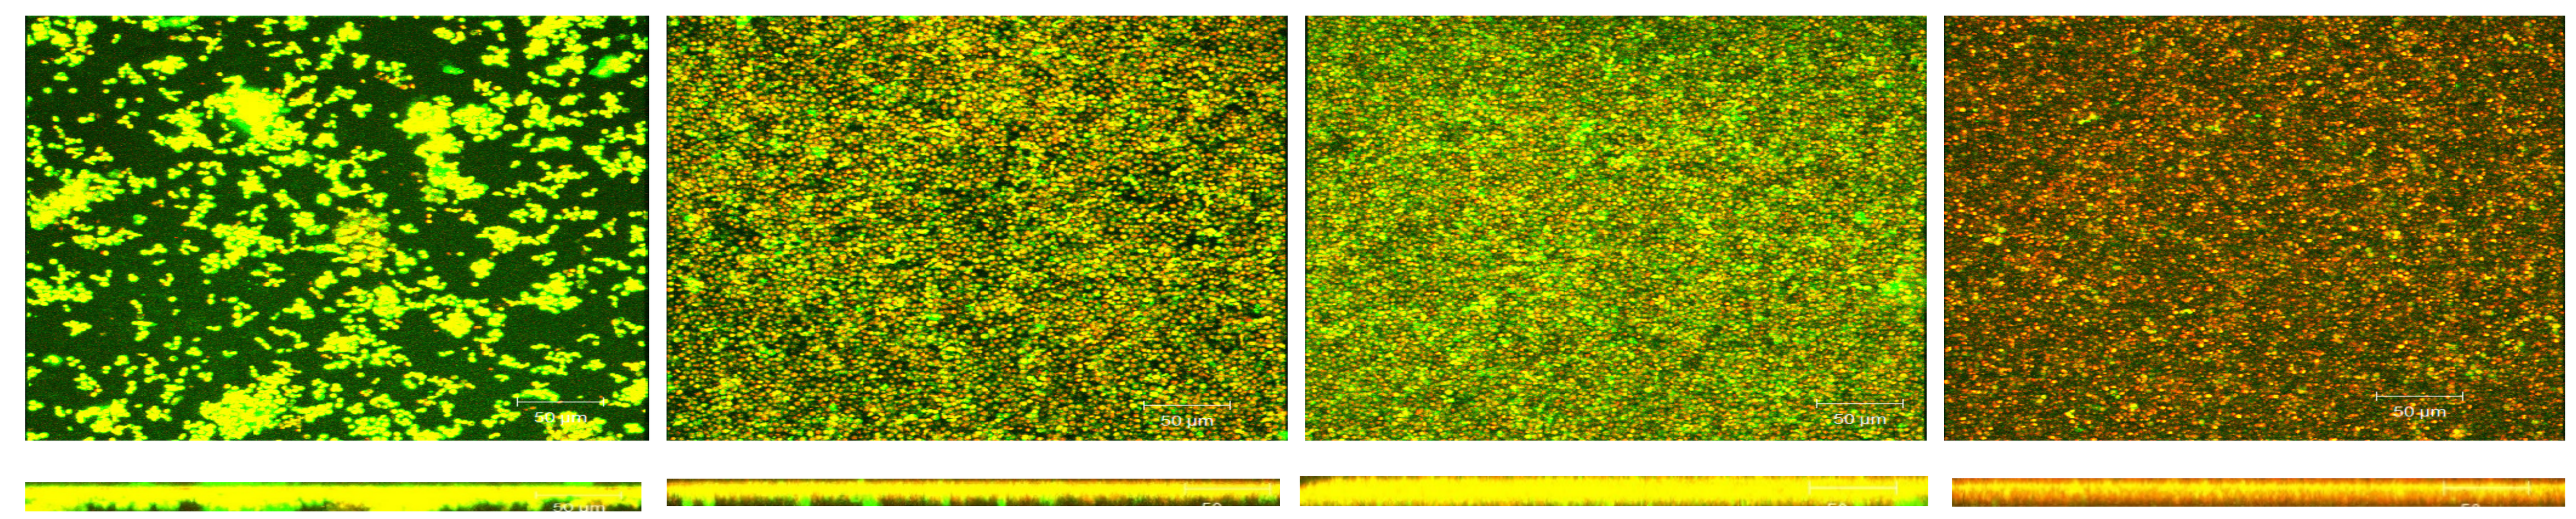

~27 um | ~16 um | ~15 um | ~15 um

CSLM top down and side views

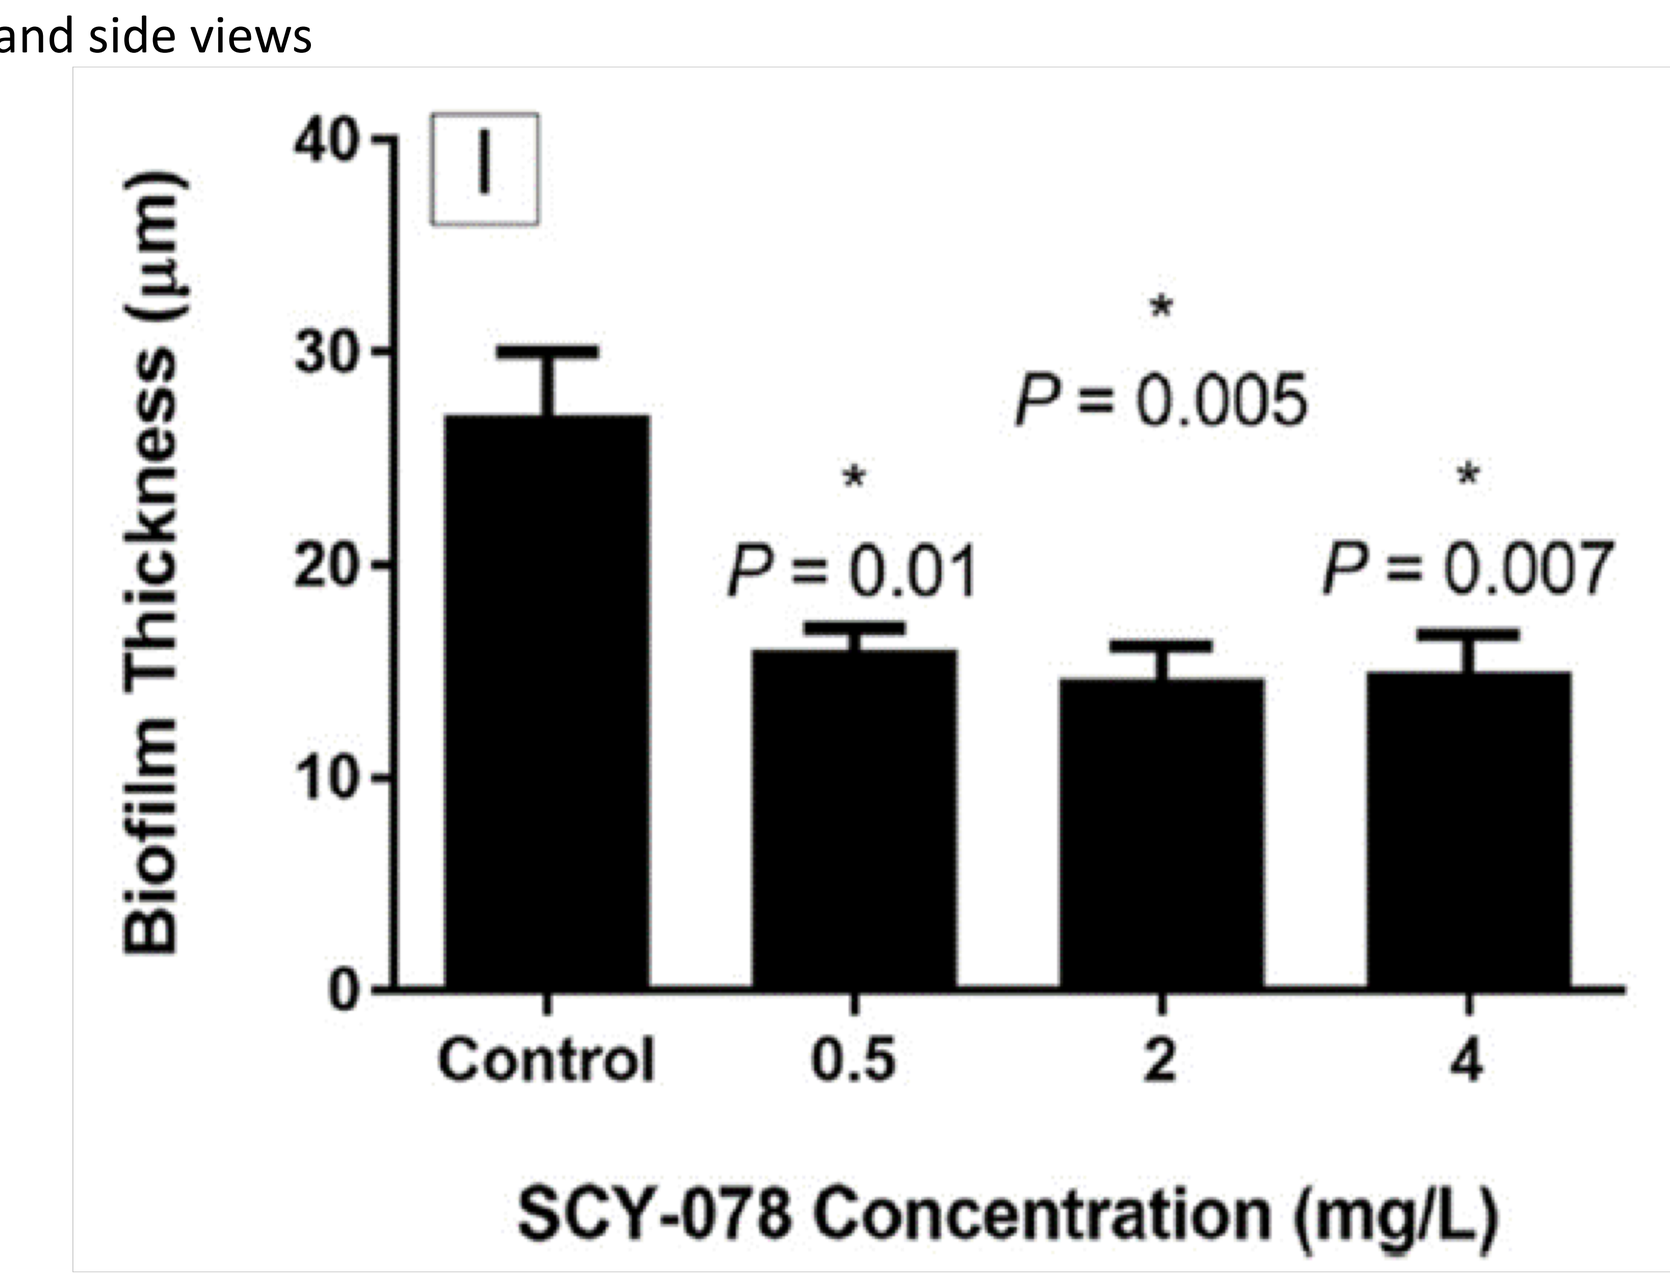
I
Biofilm Thickness (μm)
40
30
20
10
0
*
P = 0.01
*
P = 0.005
*
P = 0.007
Control
0.5
2
4
SCY-078 Concentration (mg/L)

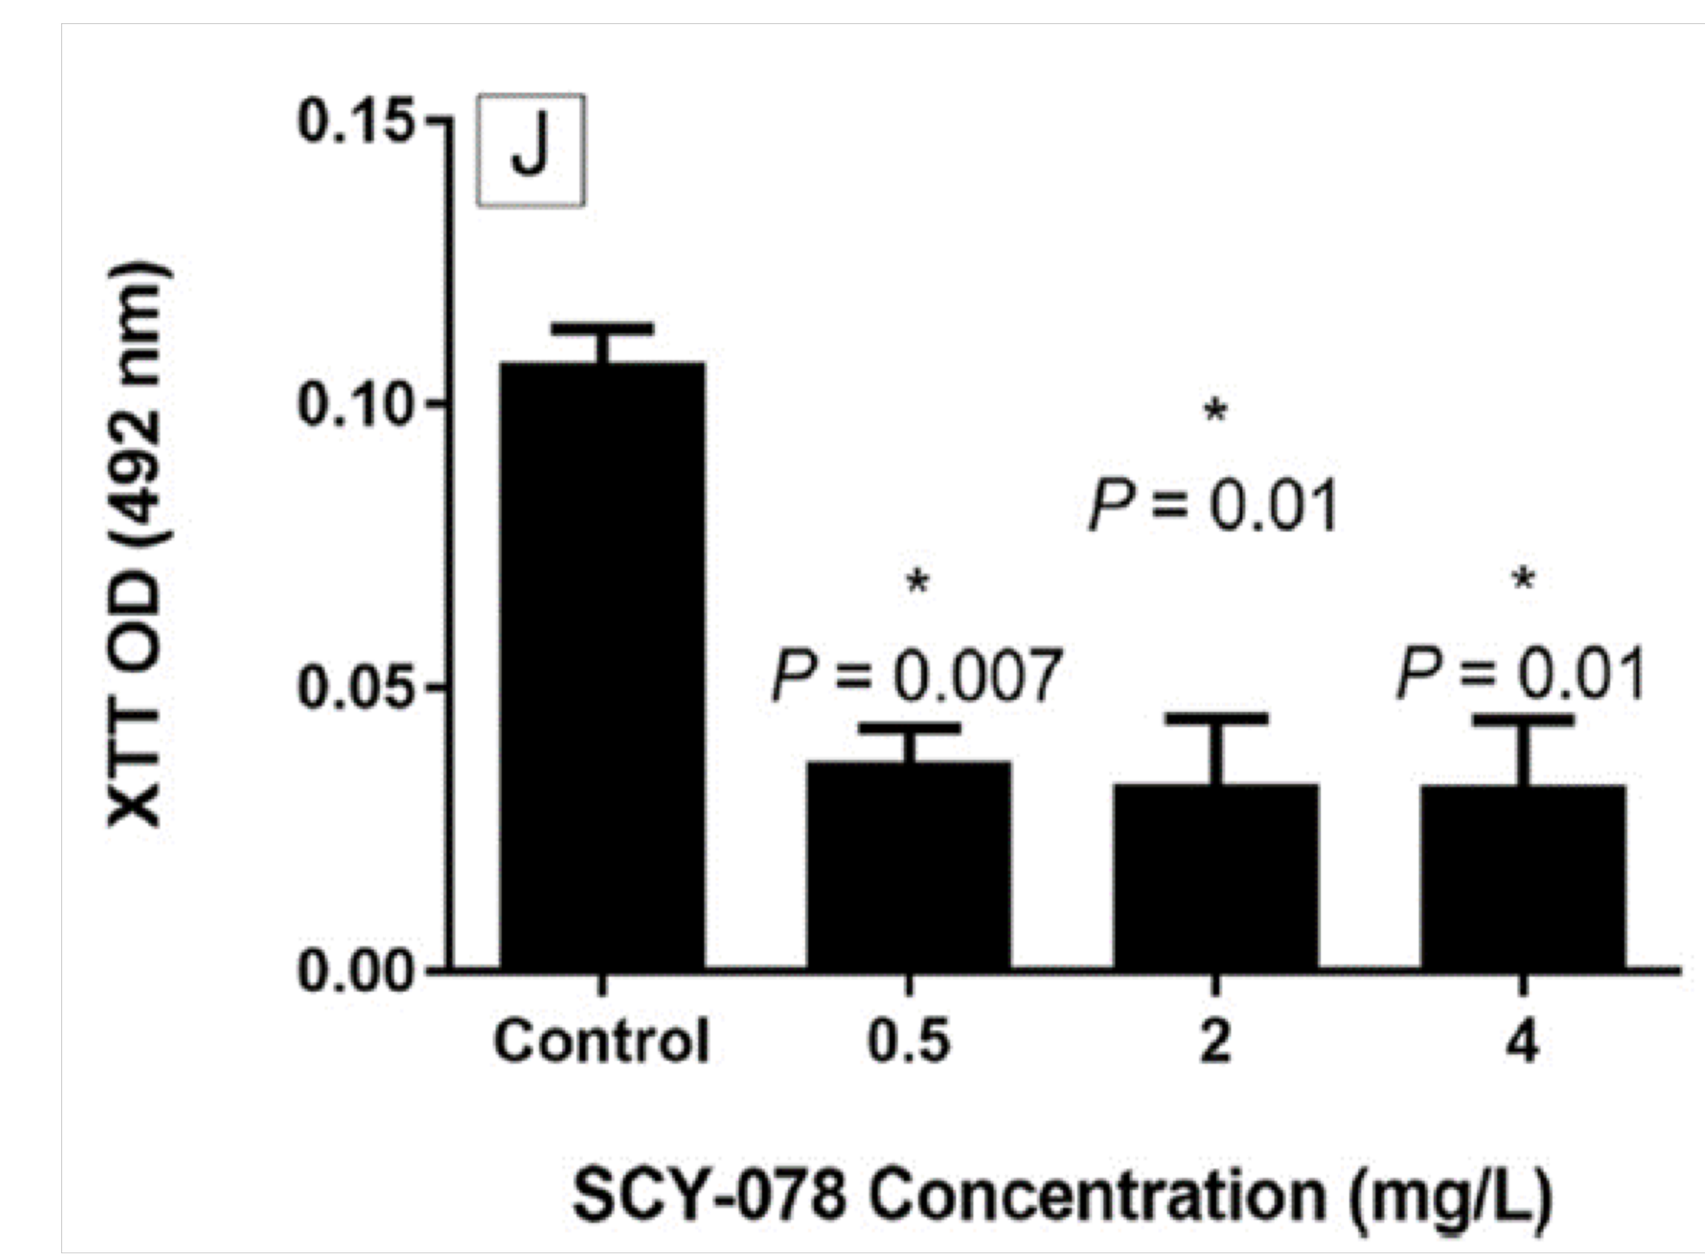
J
XTT OD (492 nm)
0.15
0.10
0.05
0.00
*
P = 0.007
*
P = 0.01
*
P = 0.01
Control
0.5
2
4
SCY-078 Concentration (mg/L)

The thickness (I) and metabolic activity (J) of untreated (control) and SCY-078-treated *C. auris* biofilms.
**P*-value compared to untreated control (P<0.05)

Transmission and scanning electron microscopy detailed the fungicidal effects of Ibrexafungerp, which consisted of abnormalities in cell division (fused cells), thickening of the cell wall with disappearance of the cell membrane, leakage of cytoplasmic matrix, and destruction of cytoplasmic organelles[a,b].

SEM

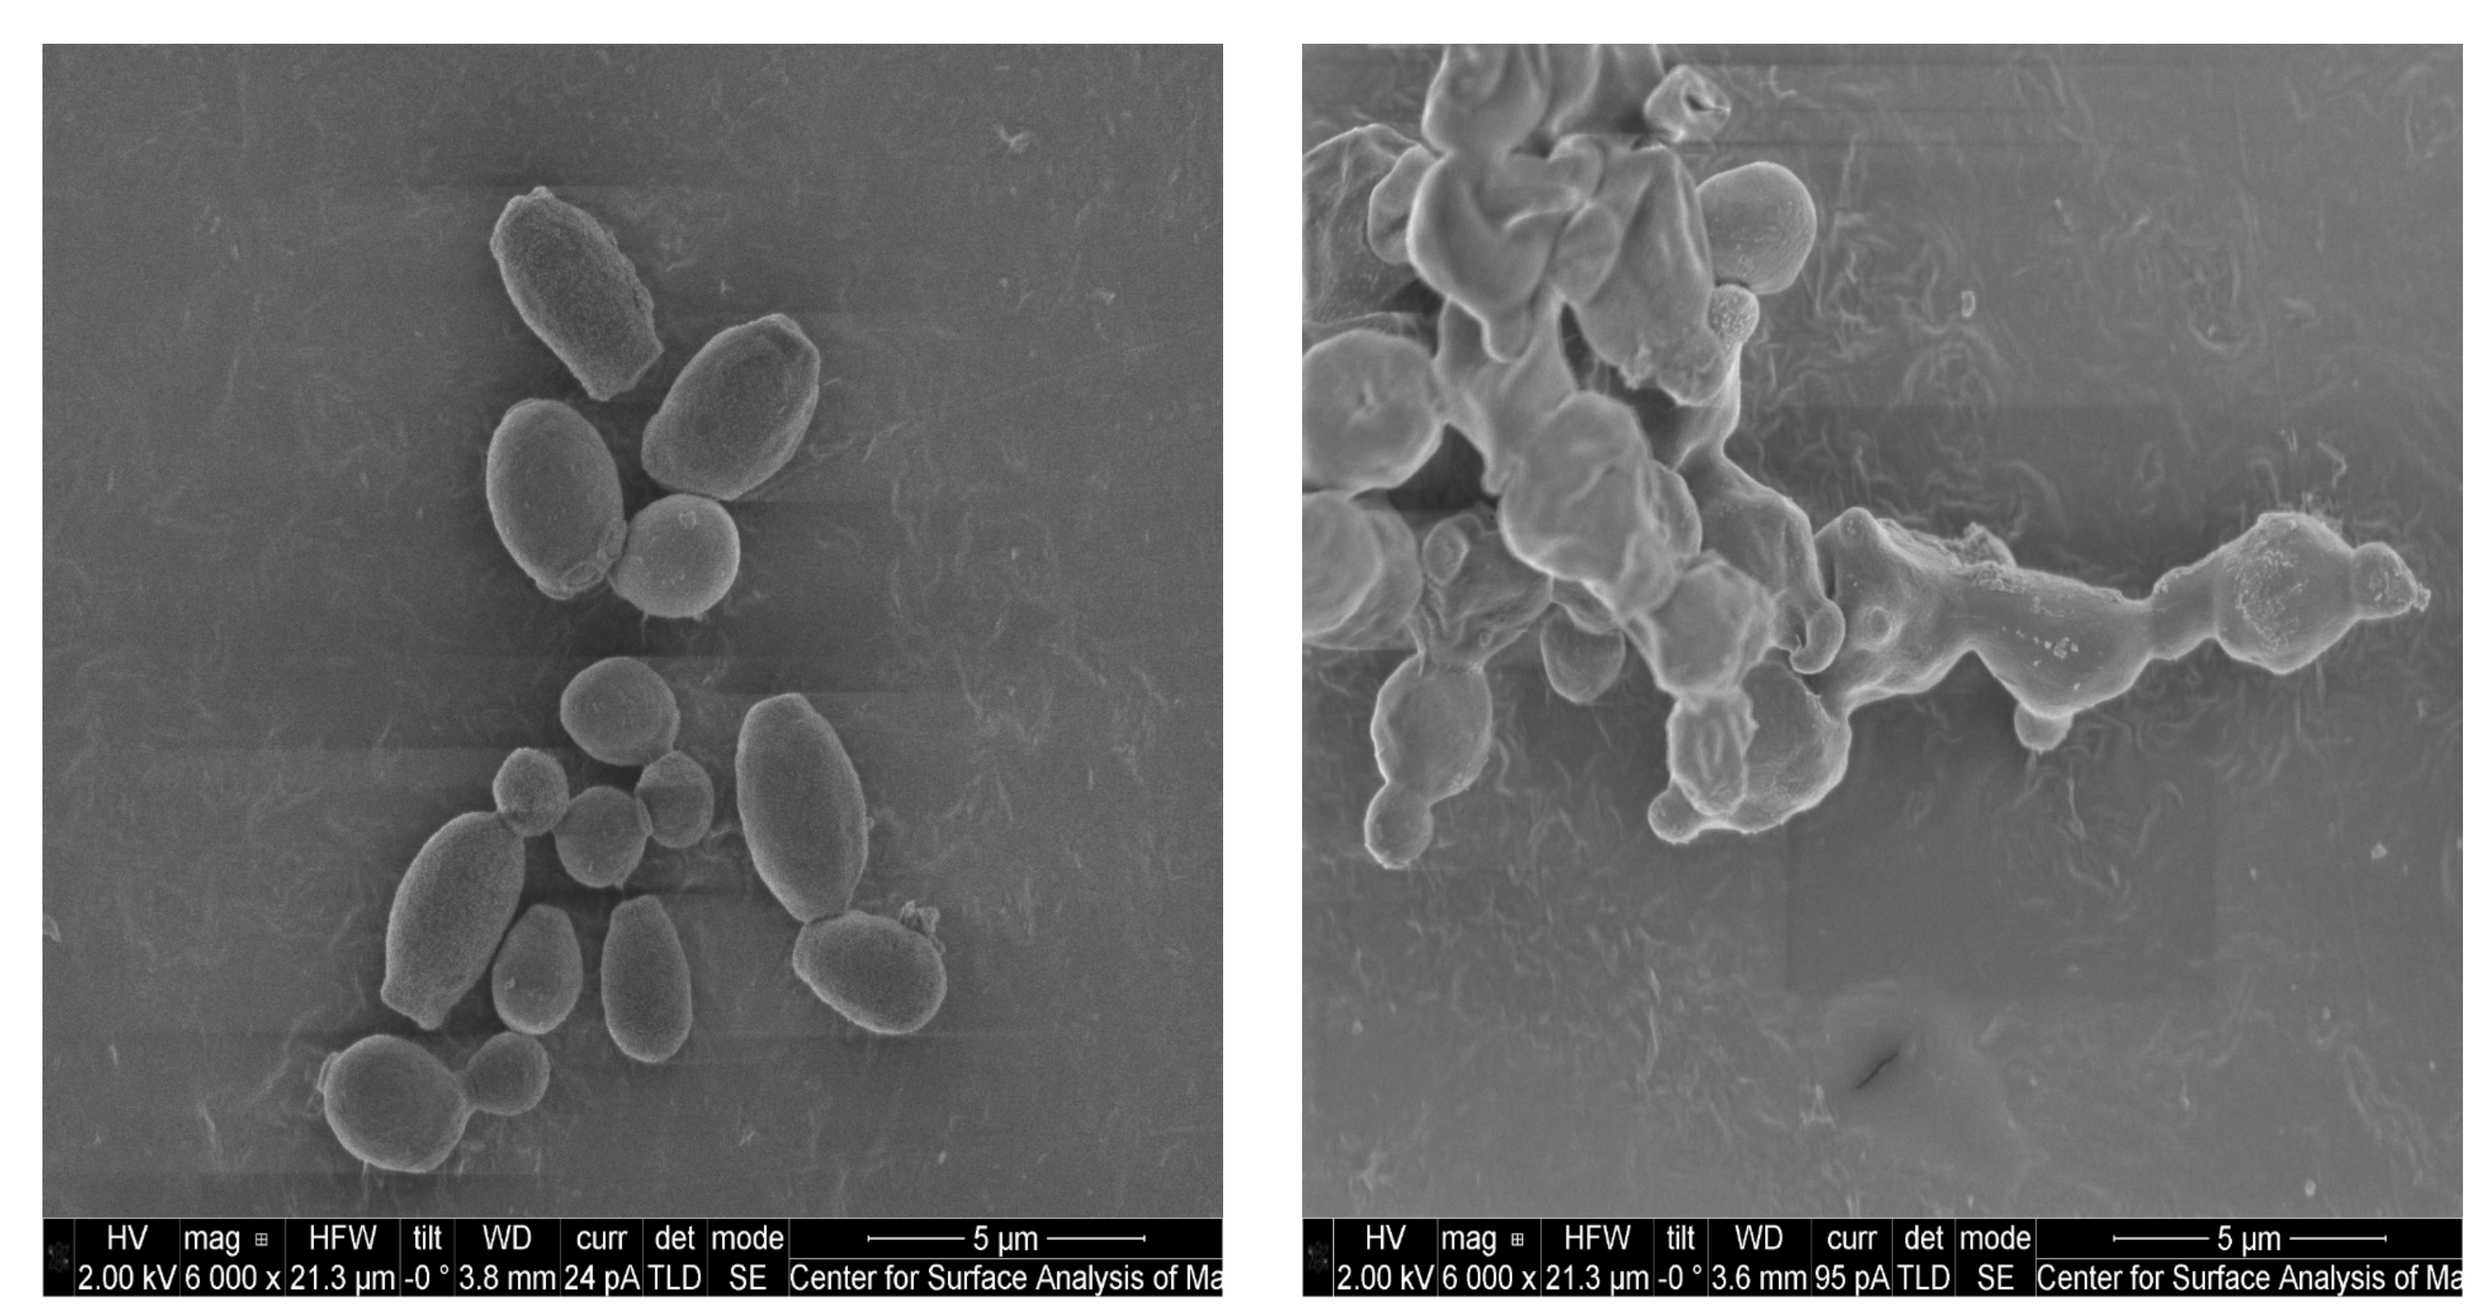
HV mag HFW tilt WD curr det mode 5 μm
2.00 kV 6 000 x 21.3 μm -0 ° 3.8 mm 24 pA TLD SE Center for Surface Analysis of Ma
HV mag HFW tilt WD curr det mode 5 μm
2.00 kV 6 000 x 21.3 μm -0 ° 3.6 mm 95 pA TLD SE Center for Surface Analysis of Ma

[a]Larkin E. et al., AAC 2017

TEM

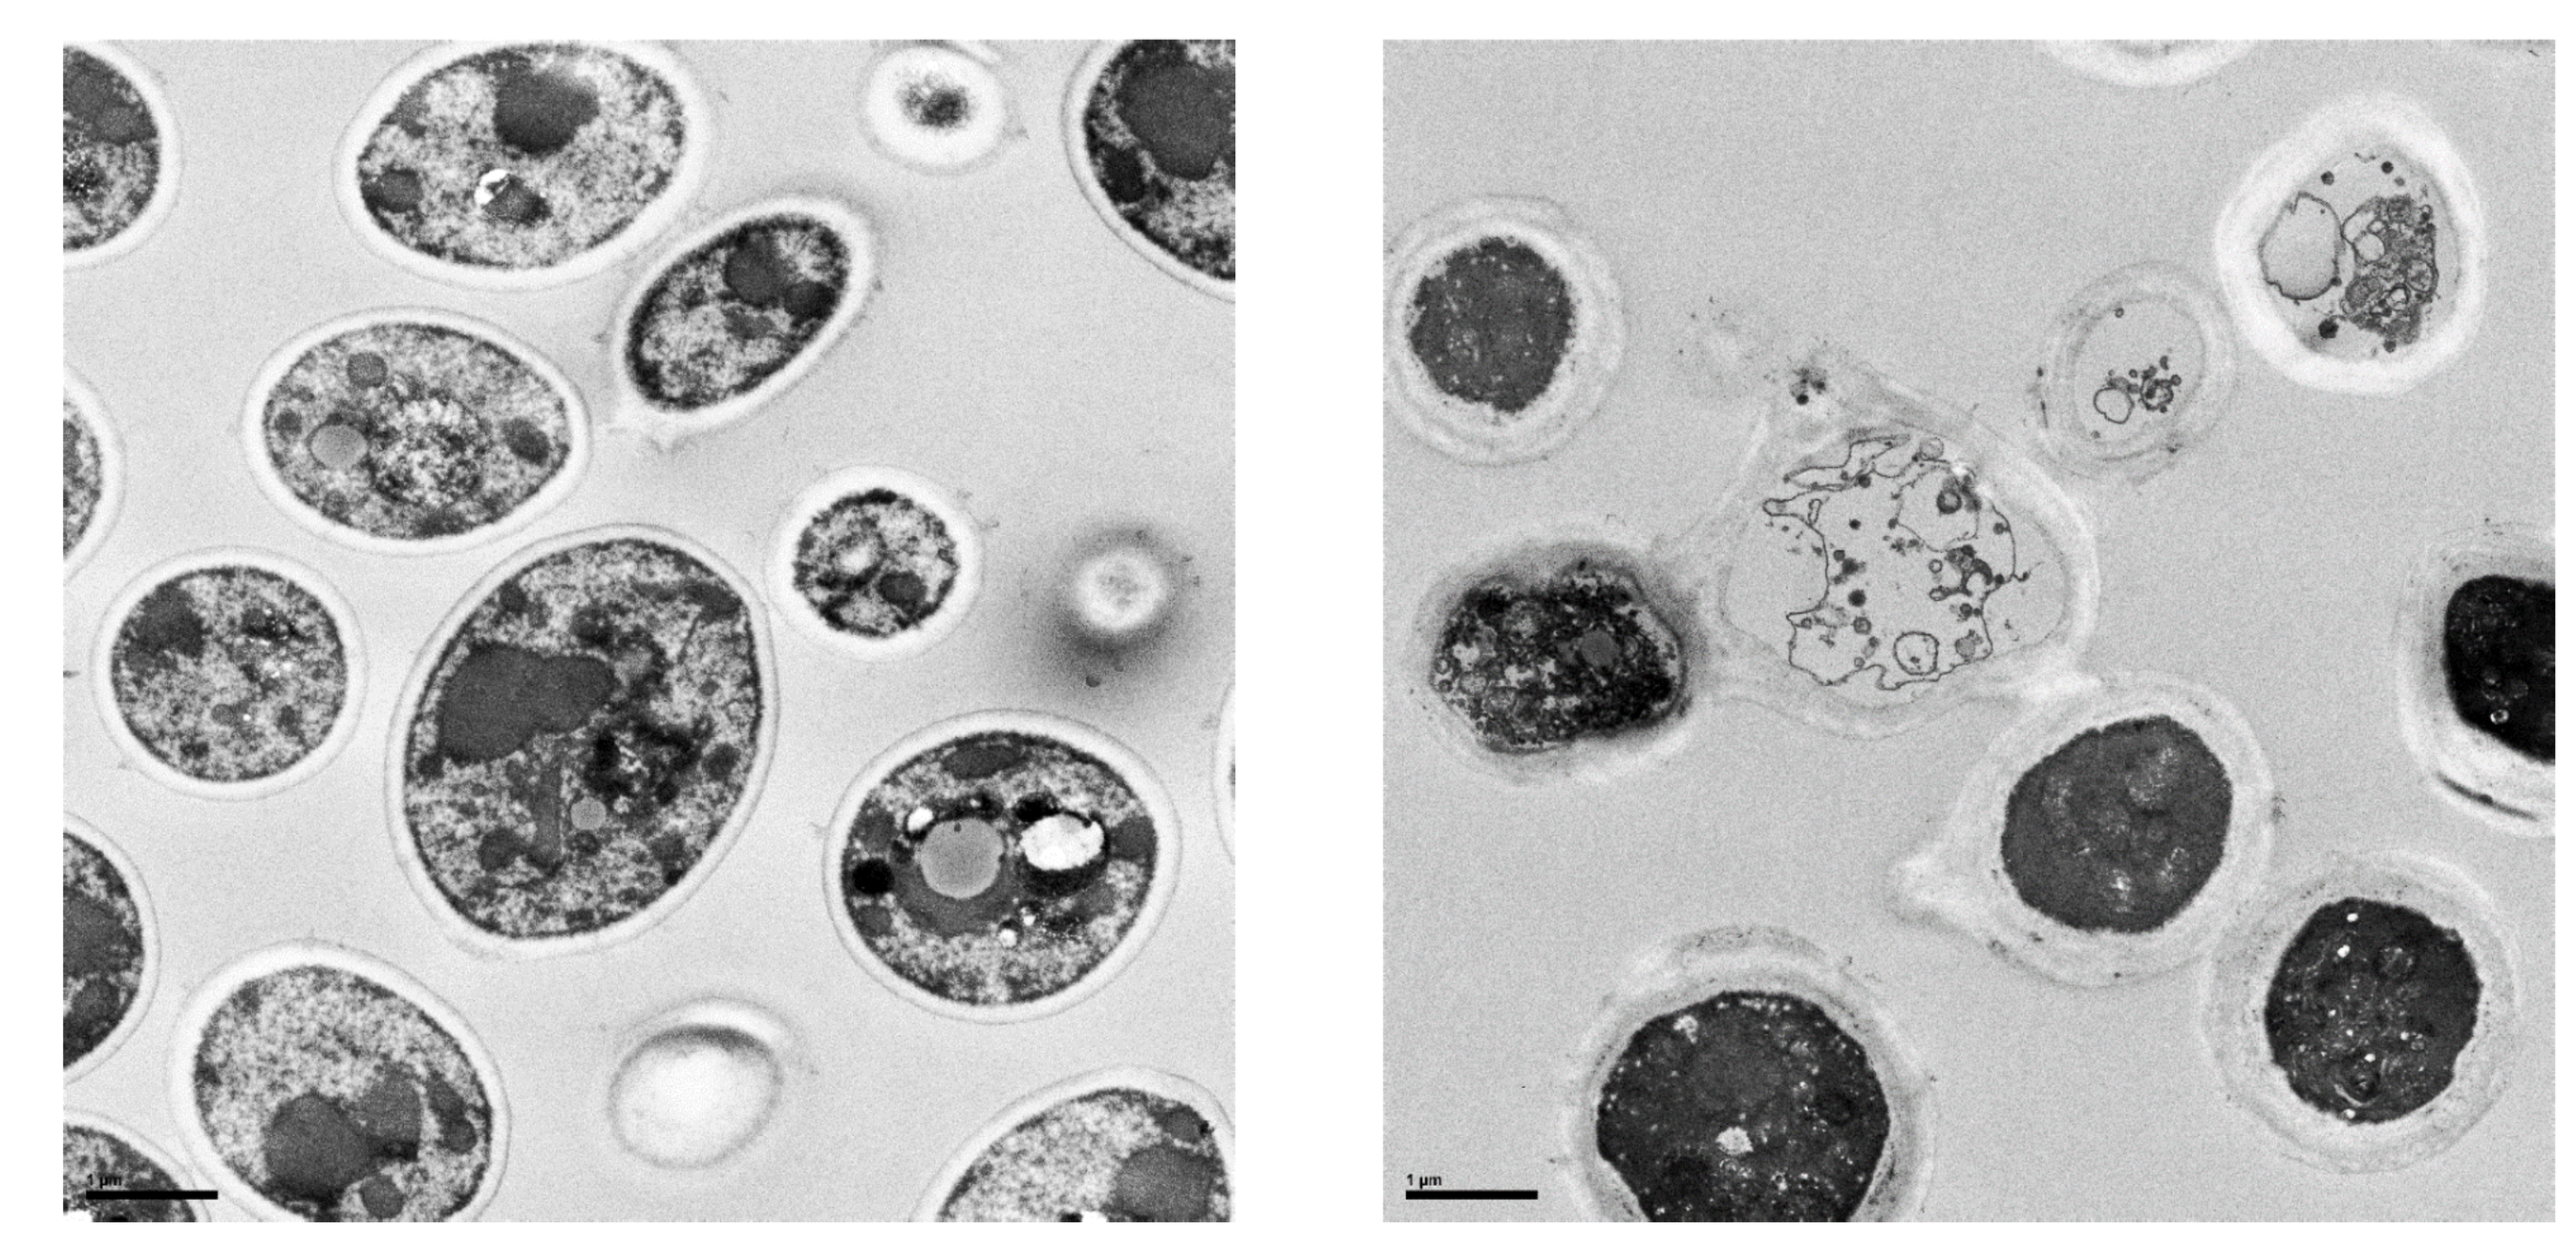

[b]Personal communications Dr. M. Ghannoum

CONCLUSIONS

Based on current *in vitro* preclinical data, Ibrexafungerp may be an important tool in the future treatment of *C. auris* infections.