The Emerging *Candida auris*: Antifungal Activity of SCY-078, a Novel Glucan Synthesis Inhibitor, on Growth, Morphology, and Biofilm Formation

E Larkin1, C Hager1, J Chandra1, PK Mukherjee1, M Retuerto1, I Salem1, L Long1, N Isham1, L Kovanda2, K Borroto-Esoda3, S Wring3, D Angulo3 and MA Ghannoum1

1. Center for Medical Mycology, Case Western Reserve University and University Hospitals Cleveland Medical Center, Cleveland, OH.
2. Astellas Pharma Global Development, Inc., Northbrook, IL. 3. SCYNEXIS, Inc., NJ, USA.

27th ECCMID Vienna, Austria 22 – 25 April 2017

INTRODUCTION

- *Candida auris* is an emerging MDR yeast species[1]
- It causes invasive infections with high mortality[1]
- It has reduced susceptibility to the three major classes of available antifungals (azoles, polyenes, and echinocandins)[1]
- Infections have been found globally
- SCY-078 is a novel IV/oral semi-synthetic triterpenoid antifungal weighing 730 Da (Fig. 1)
- MOA- inhibition of Beta 1,3 D glucan synthase inhibition;its binding properties are different than echinocandins (Fig. 2)

MATERIALS & METHODS

- Minimum inhibitory concentrations (MICs) for 11 antifungals, including SCY-078, were determined against 16 *C. auris* strains isolated in Germany, Japan, India and S. Korea using CLSI M27-A3 methodology
- Scanning electron microscopy (SEM) and confocal scanning laser microscopy (CSLM) were used to visualize the activity of SCY-078 against *C. auris* biofilms[2]
- CSLM used Fun-1 metabolic stain (converted to orange-red cylindrical intravacuolar structures by metabolically active cells) and concanavalin A-Alexa Fluor 488 conjugate (binds to cell wall polysaccharides with green fluorescence)

Triterpenoid- SCY-078 Caspofungin

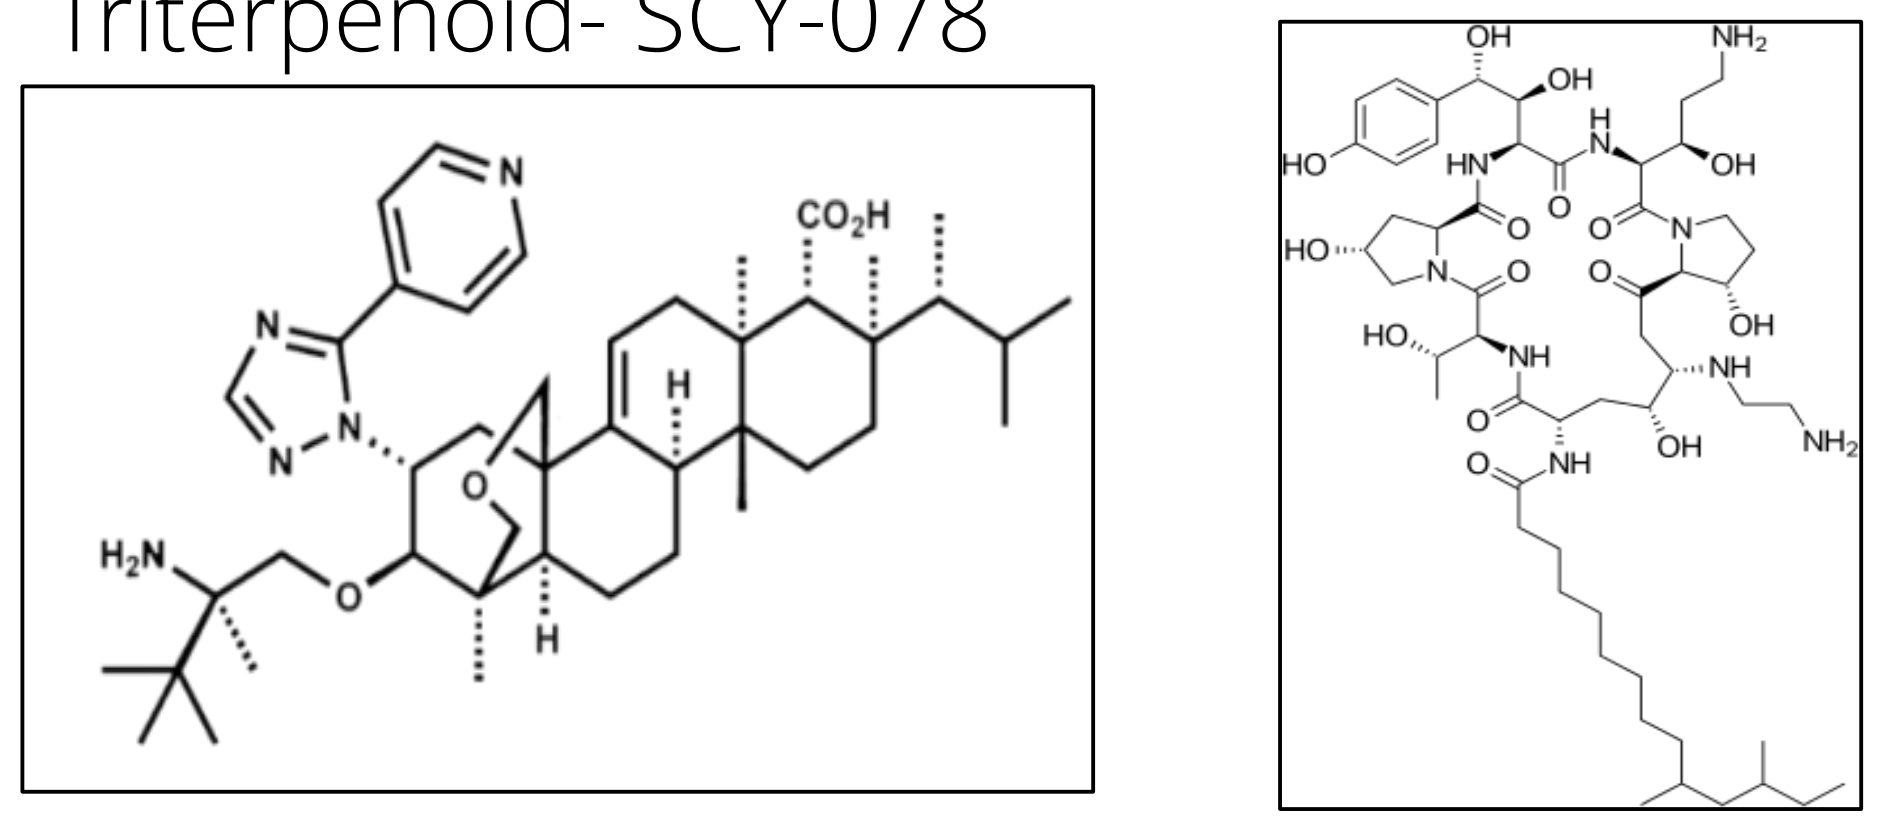

Figure 1. SCY-078 chemical structure versus Caspofungin

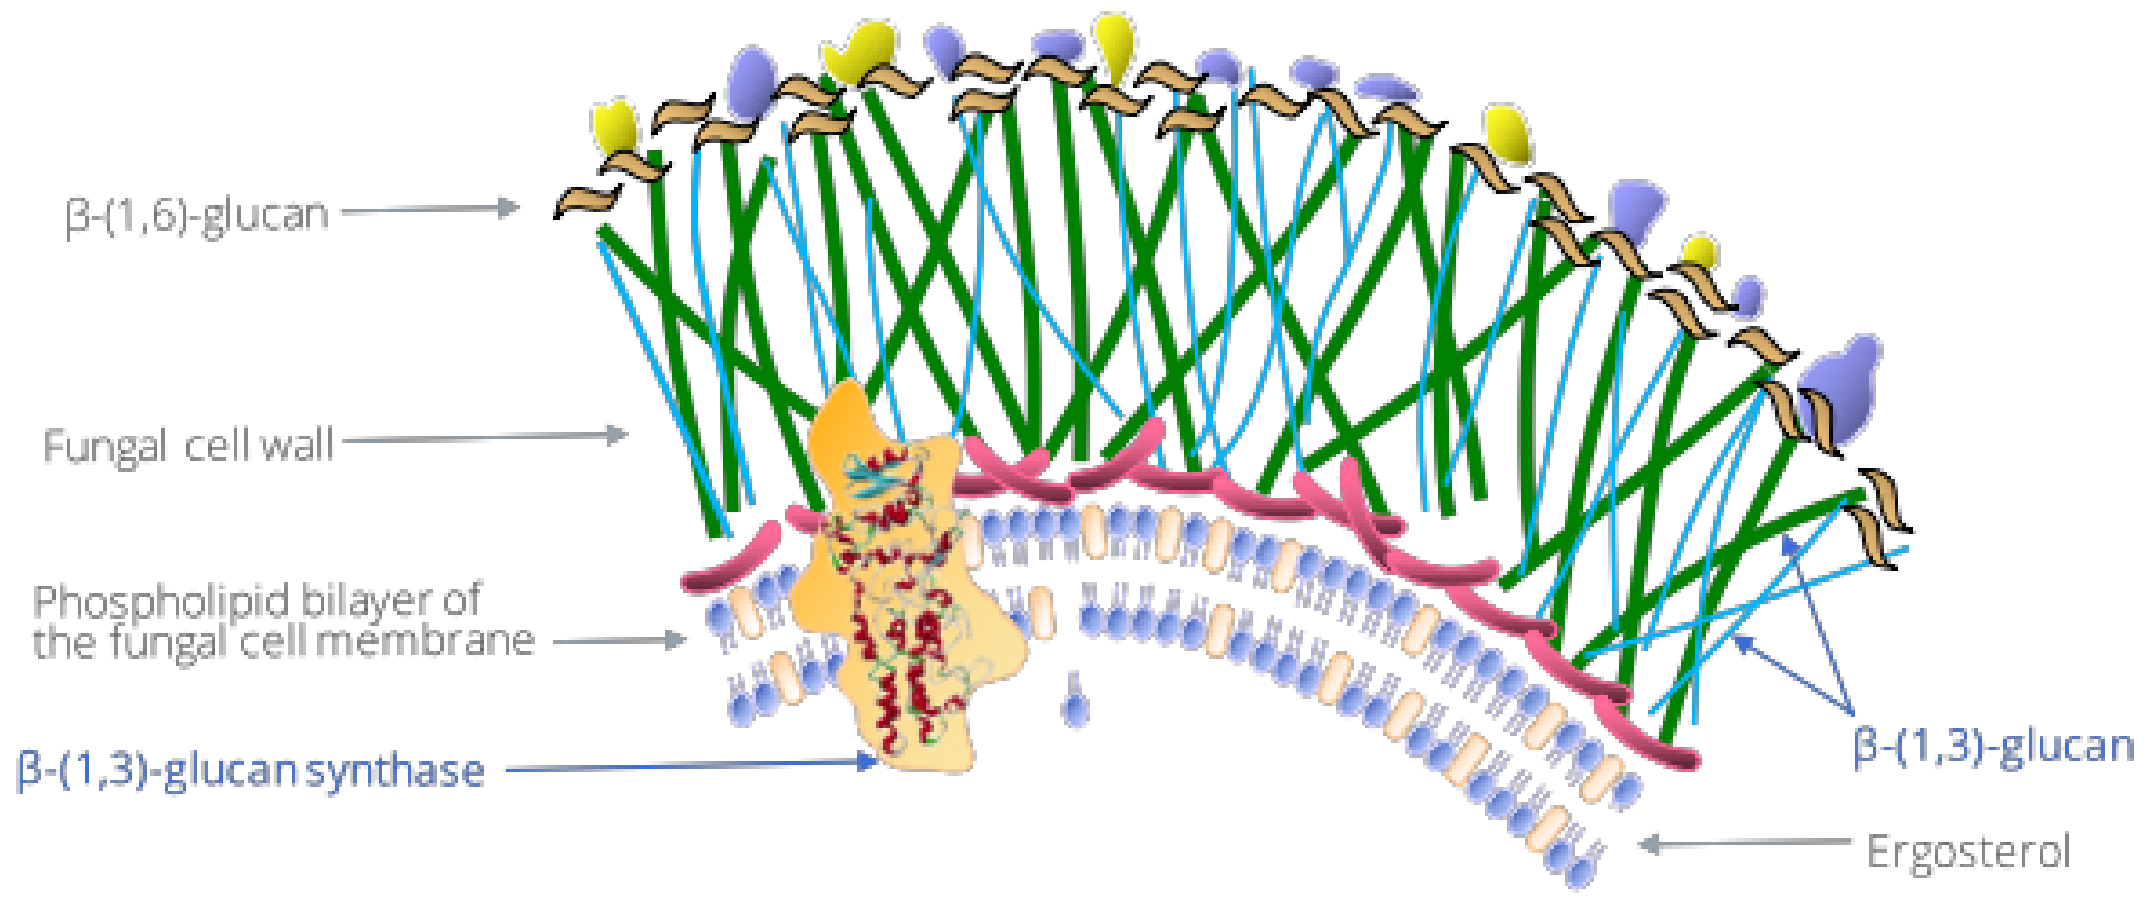
β-(1,6)-glucan
Fungal cell wall
Phospholipid bilayer of the fungal cell membrane
β-(1,3)-glucan synthase
β-(1,3)-glucan
Ergosterol

Figure 2: Fungal cell membrane, detailing Beta 1,3 D glucan synthase, SCY-078 target of inhibition

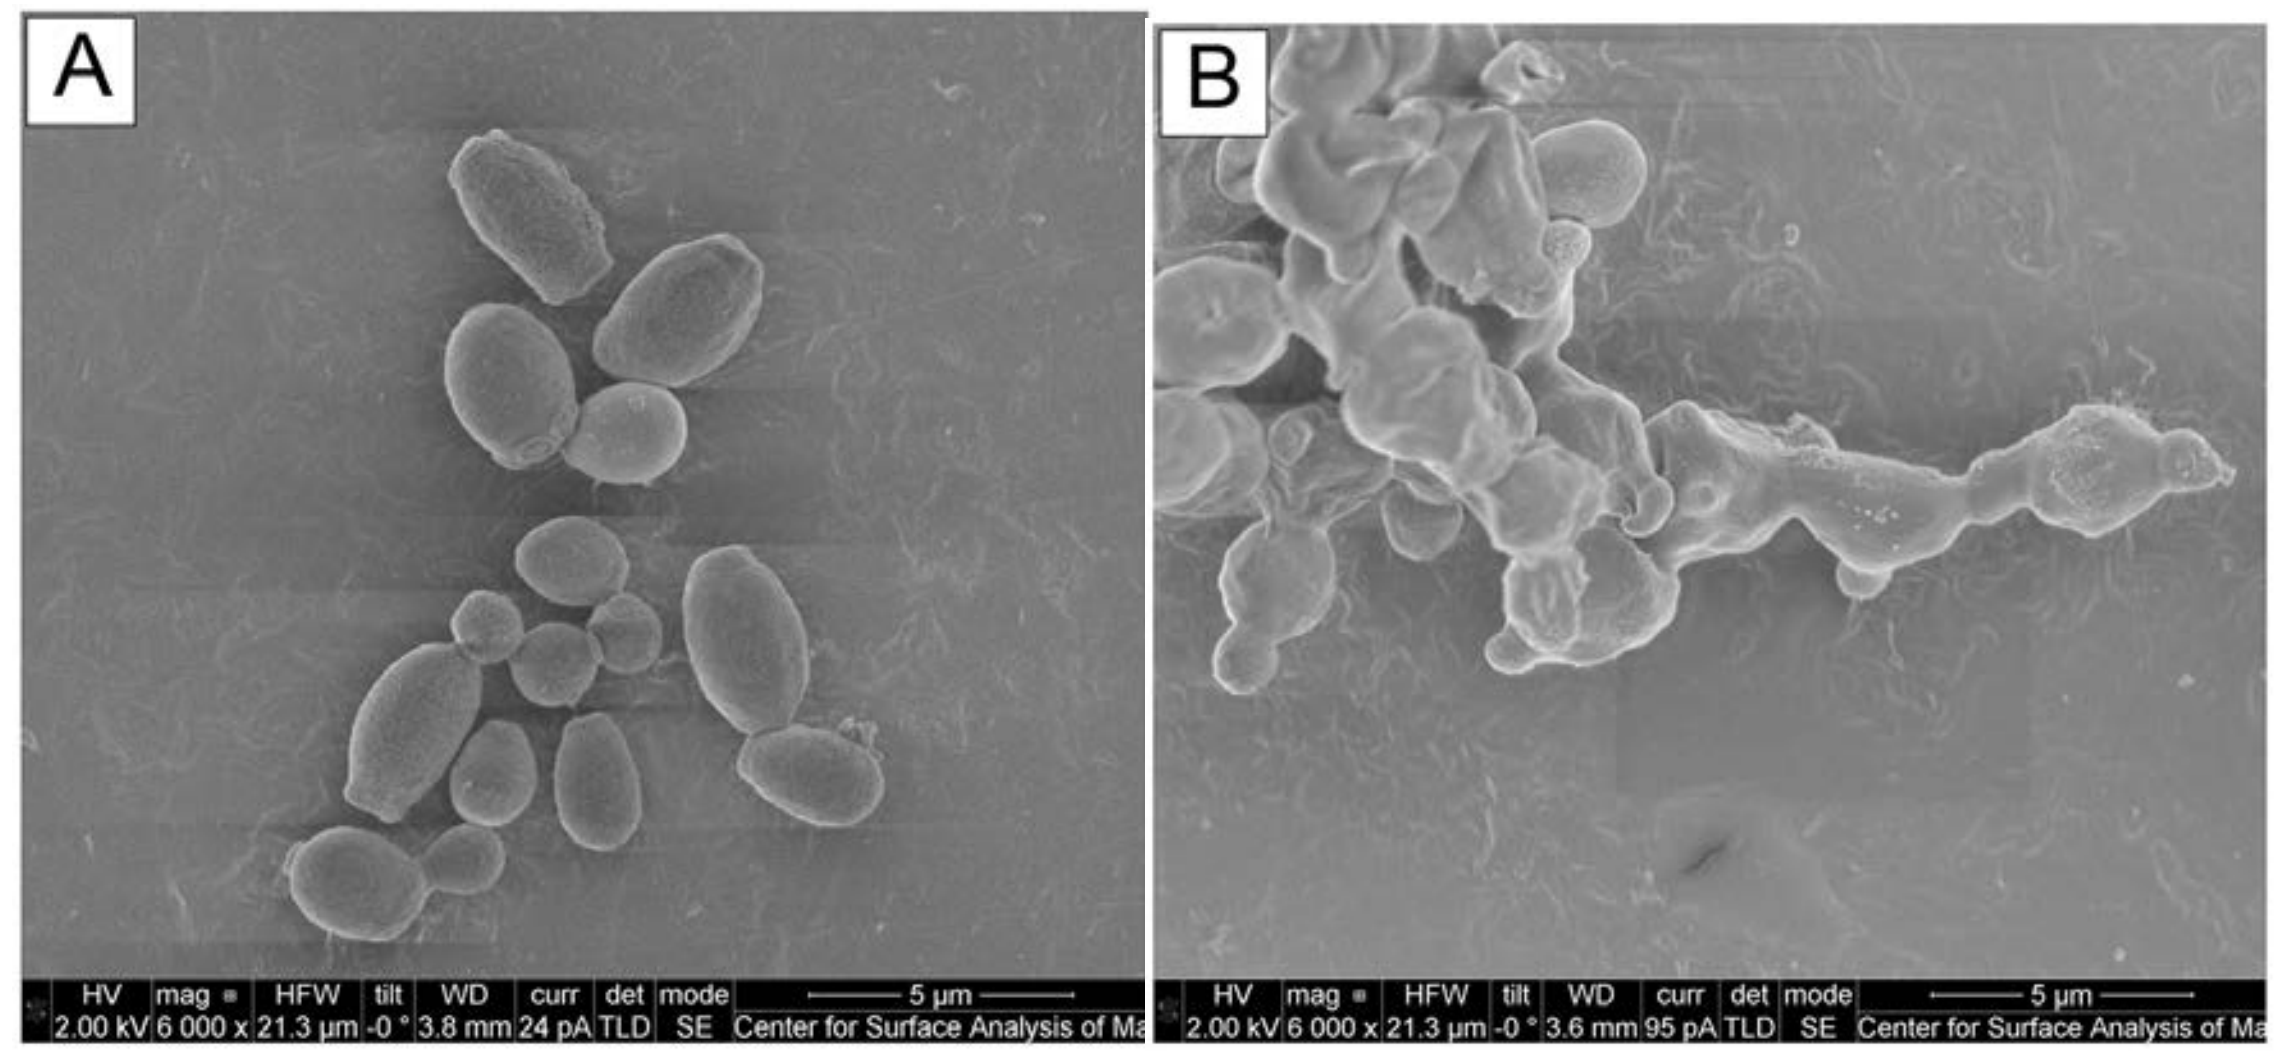
A
B
5 µm

Figure 3. Scanning electron micrograph of *C. auris* treated with: (A) no drug (control) and (B) SCY-078, at 1X MIC (0.5 mg/L)

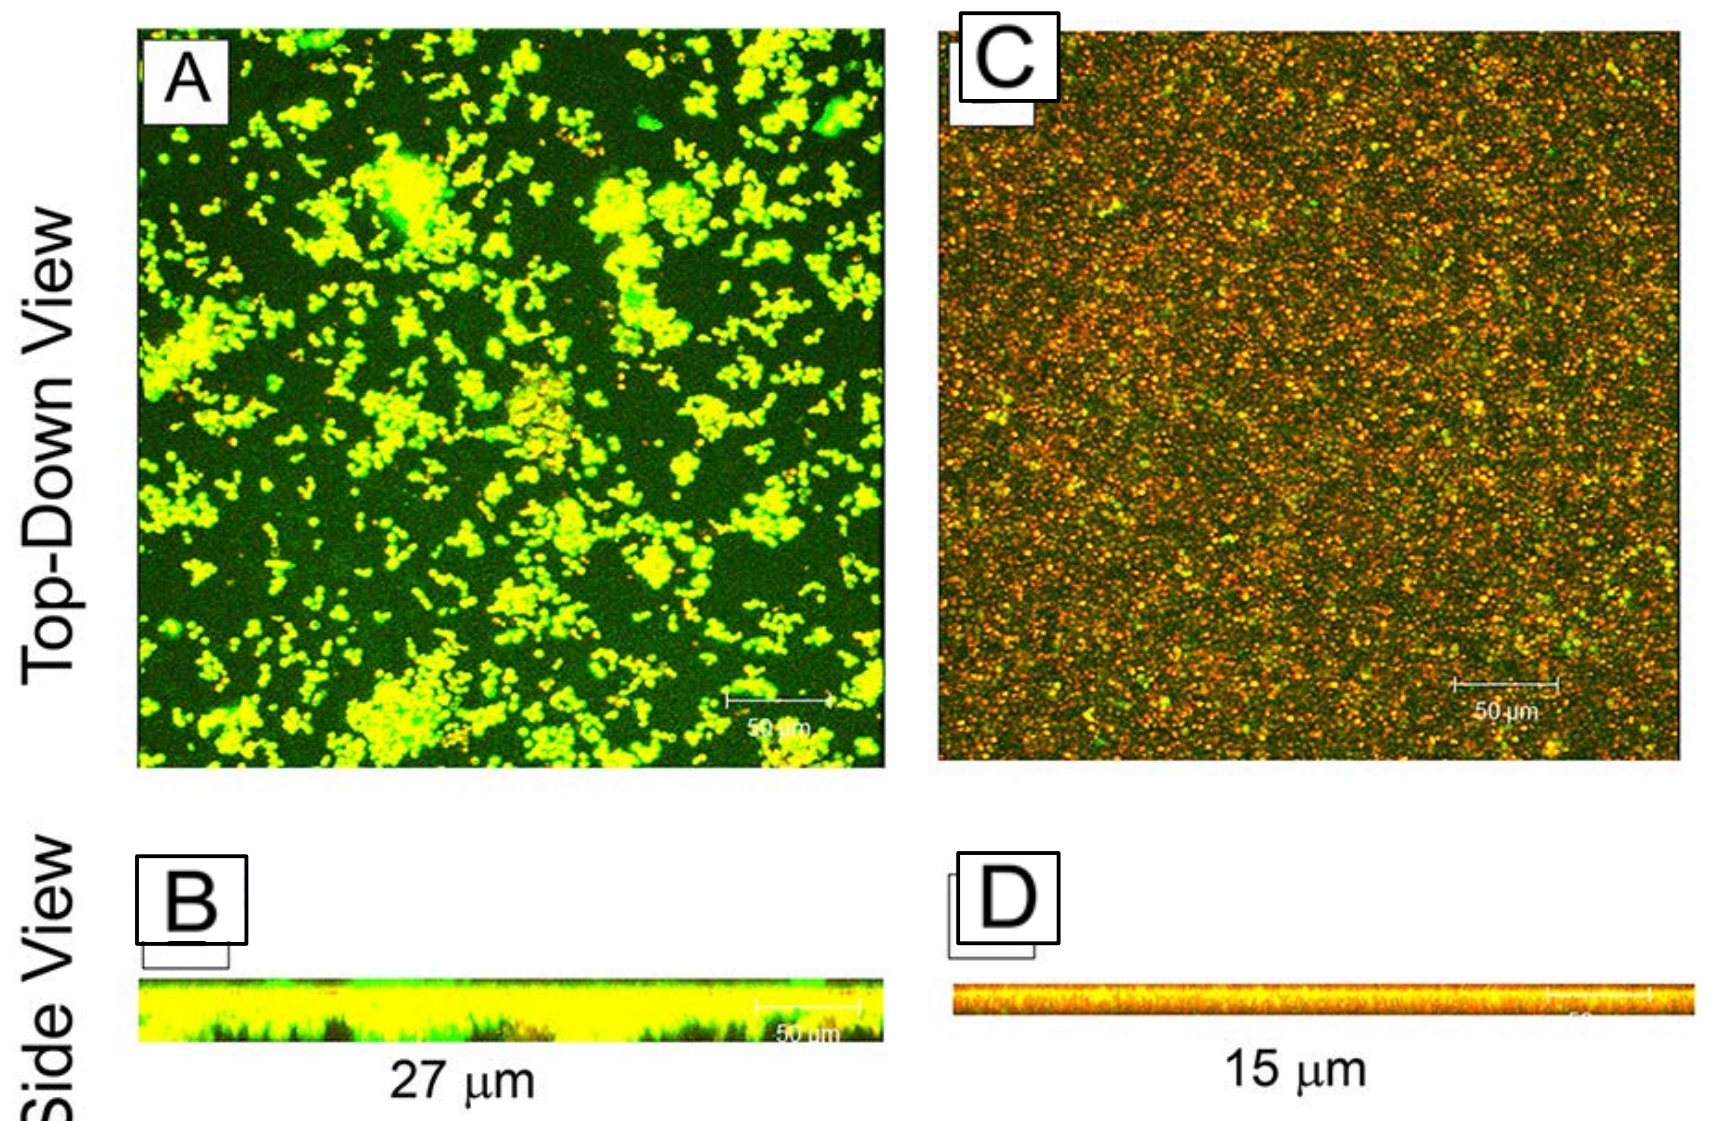
Top-Down View
Side View
A
B
C
D
27 µm
15 µm

Figure 4. Confocal scanning laser microscopy analyses of the effect of SCY-078 on biofilms formed by *C. auris* exposed to: (A, B) no drug (control) and (C, D) SCY-078, at 4 mg/L

Table 1. MIC values for SCY-078 and antifungal comparators against a panel of *C. auris* clinical isolates (n=16)

	MIC (µg/mL)										
	SCY-078	AFG	MFG	CAS	5FC	FLU	ISA	ITC	POS	VRC	AMB
Range	0.5-2	0.125-0.25	0.25-2	0.25-1	0.5-1	1->64	0.004-0.25	<0.063-1	0.25-1	<0.063-1	0.5-8
MIC_{50}	1	0.125	1	0.5	0.5	16	0.063	0.5	0.25	0.5	2
MIC_{90}	1	0.25	1	1	1	>64	0.125	1	0.5	1	4

RESULTS

- The MIC90 of SCY-078 were similar to echinocandins (1 mg/L; Table 1)
- Isavuconazole had the lowest MIC90 (0.125 mg/L) and amphotericin B and fluconazole the highest (4 and > 64 mg/L, respectively)
- SEM showed that SCY-078 interrupted *C. auris* cell division (Fig. 3)
- CSLM demonstrated reduced size of yeast cells with little or no intense fluorescence at 4 mg/L SCY-078 concentration (Fig. 4)
- SCY-078 treatment led to a reduction in biofilm thickness at all tested concentrations compared to untreated controls ($P < 0.05$)

CONCLUSIONS

- SCY-078 had potent antifungal activity against *C. auris*, with a susceptibility profile similar to that of echinocandins and activity against *C. auris* biofilm
- SCY-078, the first orally bioavailable 1,3-β-D-glucan synthase inhibitor, could be an important addition to the armamentarium against this MDR pathogen.

References

1. Calvo, B., A.S.A. Melo, A.Perozo-Mena, M. Hernandez, E.C. Francisco, F. Hagen, J.F. Meis, and A.L. Colombo. 2016. First report of *Candida auris* in America: clinical and microbiological aspects of 18 episodes of candidemia. J Infect 73:369-374.
2. Clinical and Laboratory Standards Institute. 2008. Reference method for broth dilution antifungal susceptibility testing of yeasts--third edition: approved standard M27-A3, vol. CLSI, Wayne, PA, USA.

EP0698

For additional information contact Mahmoud A. Ghannoum, PhD at mag3@case.edu or SCYNEXIS at info@scynexis.com.