RX-3117, A NOVEL, ORAL NUCLEOSIDE ANALOG, IN COMBINATION WITH NAB-PACLITAXEL (NAB-PAC) IN FIRST-LINE METASTATIC PANCREATIC CANCER (MET-PC): SAFETY, TOLERABILITY AND PRELIMINARY RESPONSES

BABIKER H[1], SCHLEGEL P[2], BENAIM E[3], HEATON CM[3], PETERSON C[3], OCEAN A[4]

1 University of Arizona Cancer Center, Tucson, AZ, USA; 2 Cancer Care Northwest, Spokane, WA, USA; 3 Rexahn Pharmaceuticals, Inc., Rockville, MD, USA ; 4 New York Presbyterian-Weill Cornell Medical Center, New York, NY, USA

Abstract

Background: RX-3117 is an oral small molecule antimetabolite that is activated by uridine cytidine kinase 2 (UCK2) which is predominantly expressed in cancer cells. RX-3117 has demonstrated safety and efficacy in a phase 1b/2a single agent clinical study in previously treated patients with met-PC or advanced bladder cancer. RX-3117 in combination with nab-pac is being evaluated for the 1st line treatment of met-PC cancer.

Methods: This study (NCT03189914) is a multicenter, open label, single arm study, conducted in 2 phases. Eligible subjects (aged ≥ 18 years) have histologically or radiologically proven met-PC with no prior therapies for metastatic disease, ECOG PS 0-1, and normal organ function (hematologic, renal, and liver). Subjects receive the MTD of single agent RX-3117, (Adashek, J, (2018) JCO; 36:4543 (abstr)) (700 mg administered orally once-daily for 5 consecutive days with 2 days off per week) and standard nab-pac (125 mg/m^2 administered IV once weekly) for 3 weeks with 1 week off per 4-week cycle in a dose de-escalation design. The primary endpoints of Phase 1 are the safety of the combination in met-PC and to determine the target Phase 2 dose. The primary endpoint of Phase 2 is PFS and/or objective clinical response.

Results: As of August 30, 2018, 8 Phase 1 subjects and 13 Phase 2 subjects were enrolled and treated (9 males and 12 females, median age 67 years). The target Phase 2 dose was 700 mg RX-3117 administered orally once-daily for 5 consecutive days with 2 days off per week) and nab-pac (125 mg/m^2 administered once weekly) for 3 weeks with 1 week off per 4-week cycle. The most common (≥20%) adverse events were diarrhea, nausea, fatigue, alopecia, anorexia, and rash. No DLTs were reported. Eleven subjects had at least one on-study scan (after 2 cycles). One subject experienced a complete response (CR) after 6 cycles of therapy with normalization of CA19.9 (-76%).Three subjects exhibited a partial response (PR): two after 2 cycles (39-47%) and one after 4 cycles of therapy (36%). Five subjects experienced stable disease for at least 2 cycles with tumor marker reductions of 43 – 69%. In evaluable patients the disease control rate (CR+PR+SD) was 82%. Overall response rate (CR+PR) was 36%.

Conclusions: RX-3117 in combination with nab-pac at the target dose appear to be safe and well-tolerated in patients with met-PC. Early anti-tumor activity is seen. Phase 2 of the clinical study is ongoing.

Introduction

The current National Comprehensive Cancer Network recommendations suggest acceptable chemotherapy combinations for met-PC patients include FOLFIRINOX, gemcitabine plus nab-paclitaxel (nab-pac), and gemcitabine plus erlotinib. However, clinical trials are also recommended due to the lack of efficacy and severe toxicity (FOLFIRINOX) with these treatments. RX-3117, an oral small molecule antimetabolite, is phosphorylated by uridine-cytidine kinase 2 (UCK2), an enzyme found in cancer cells, but relatively absent in normal cells, allowing a targeted approach. The targeted approach may allow for maximum dosing of RX-3117 and agents used in combination, such as nab-pac, without compromising safety. Additionally, RX-3117 is inactivated by cytidine deaminase at a slow rate, allowing higher cellular concentrations and an increased likelihood of antitumor activity at the appropriate stage of the cell cycle. Efficacy and safety of RX-3117 as a single agent in metastatic pancreatic cancer has been previously reported (Adashek, J, (2018) JCO; 36:4543 (abstr).

Study Design

This is a multicenter, open-label, 2-stage Phase 1b/2a study. Phase 1 was a dose-finding, open-label study of oral RX-3117 administered in combination with nab-pac to subjects with newly diagnosed met-PC. The recommended phase 2 dose (RP2D) of RX-3117, in combination with nab-pac, was determined by the safety profile and dose modification to be 700 mg administered orally once-daily for 5 consecutive days with 2 days off per week and nab-pac 125 mg/m^2 administered IV once weekly for 3 weeks with 1 week off per 4-week cycle. After completion of the Phase 1 portion, the Phase 2 study began using a 2 stage, open-label design, of RX-3117 and nab-pac in combination to treat subjects with met-PC as first line therapy. In the Stage 1 approximately 10 subjects participated at the dose identified in the Phase 1 (RP2D). Subjects will be treated for up to 8 cycles of combined therapy. The protocol provided for an interim analysis to be performed after the first 10 evaluable subjects in Stage 1 were treated for a minimum of 4 cycles. However, since two PRs were already achieved in the first seven subjects, the study met the criteria for proceeding to Stage 2. Approximately 30 additional evaluable subjects will be enrolled to participate in Stage 2 (dose expansion). Subjects will be treated for up to 8 cycles of combined therapy.

Demographics

Gender	n (%)	Age	Median (Range)
Male	9 (47.4%)	67 (45-81)	
Female	10 (52.6%)		
Race	**n (%)**	**ECOG**	**n (%)**
White	18 (94.7%)	0	8 (42.1%)
Black or African American	1 (5.3%)	1	11 (57.9%)

Note: Data from Phase 1 and Stage 1 of the Phase 2.

Safety

Grade 3-4 Adverse Events by Relationship

TEAE	Unrelated	nab-pac	RX-3117	Both	Total n (%)
Decreased appetite	0	1	0	2	3 (15.8%)
Diarrhea	0	0	0	3	3 (15.8%)
Fatigue	0	0	0	3	3 (15.8%)
Anemia	1	0	0	1	2 (10.5%)
Dehydration	0	1	0	1	2 (10.5%)
Febrile neutropenia	0	2	0	0	2 (10.5%)
Nausea	0	0	0	2	2 (10.5%)
Neutropenia	1	0	0	1	2 (10.5%)
Urinary tract infection	1	0	0	1	2 (10.5%)
White blood cell count decreased	0	1	0	1	2 (10.5%)

- 8 subjects were treated and 6 were evaluable in Phase 1. No DLTs occurred.
- Most Treatment Emergent Adverse Events (TEAEs) considered related to RX-3117, nab-pac, or the combination were mild to moderate (82%). The most common mild to moderate TEAEs related to RX-3117, nab-pac, or the combination were nausea, diarrhea, fatigue, alopecia, decreased appetite, rash, pruritus, pyrexia, and vomiting.

Results

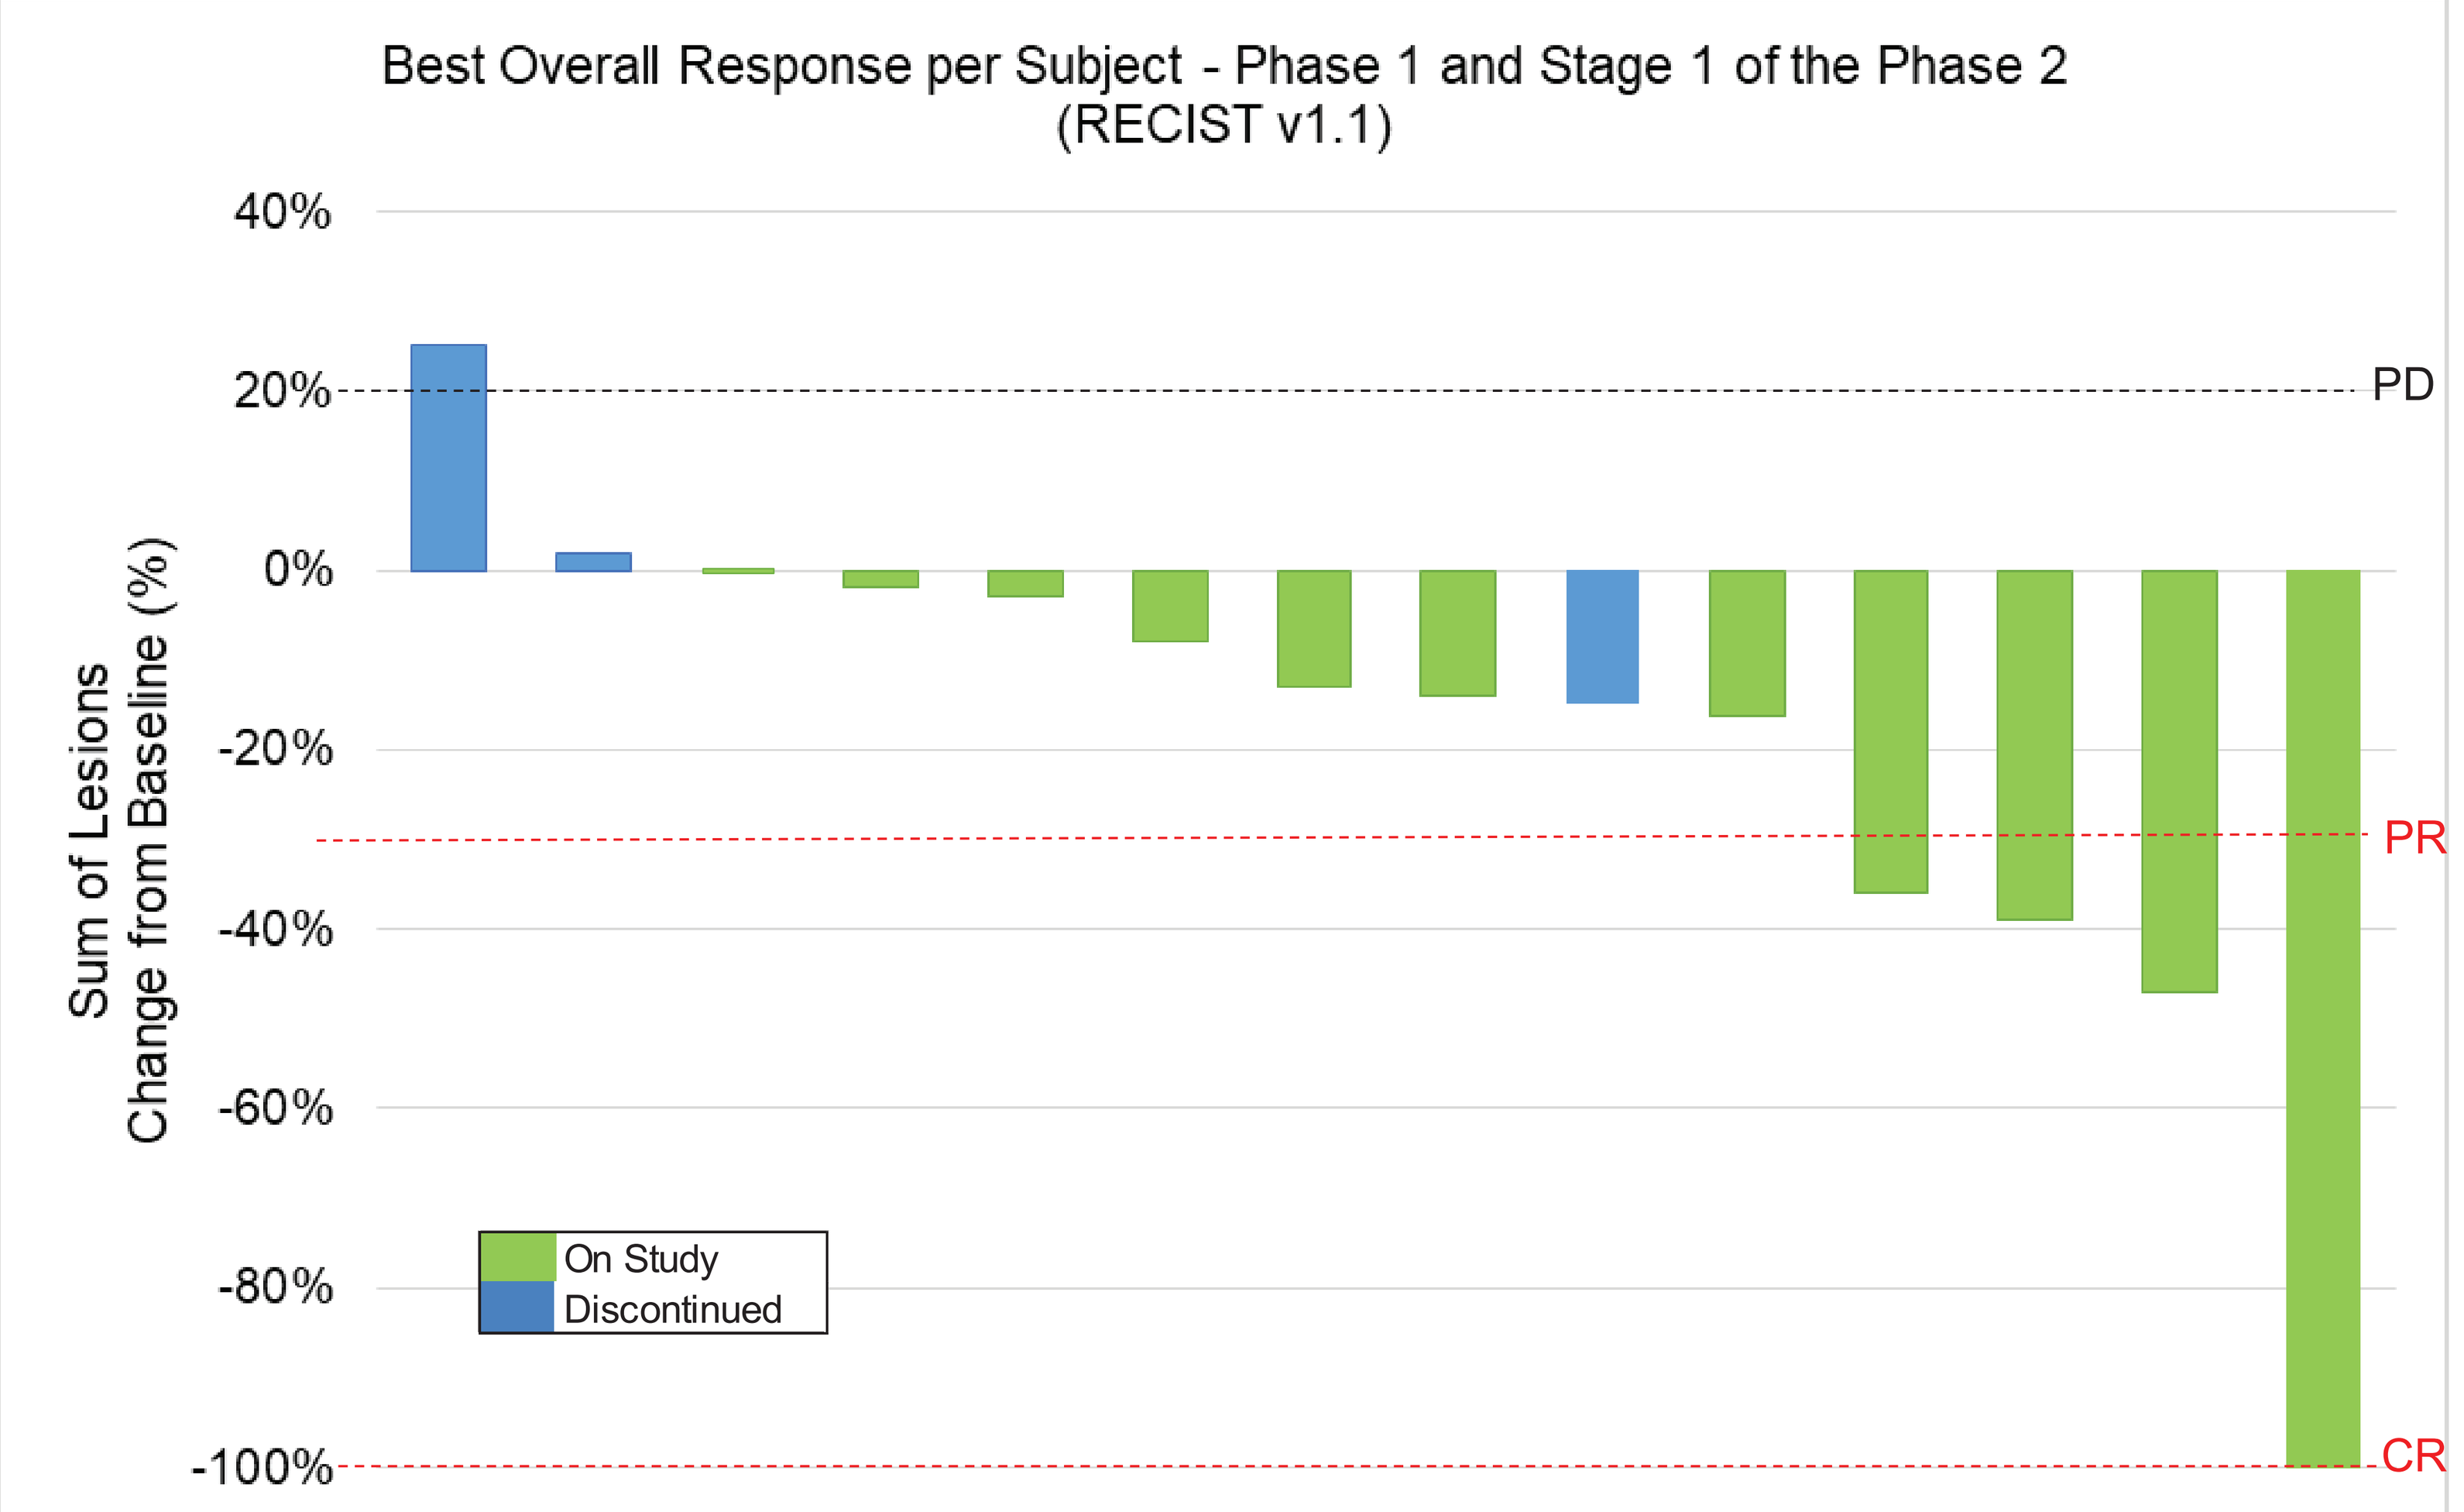
Best Overall Response per Subject - Phase 1 and Stage 1 of the Phase 2
(RECIST v1.1)
Sum of Lesions
Change from Baseline (%)
40%
20%
0%
-20%
-40%
-60%
-80%
-100%
PD
PR
CR
On Study
Discontinued

Note: 13 subjects had at least one post-treatment scan and were response evaluable. Two (out of 7) Phase 2 – Stage 1 subjects had partial responses. Three subjects discontinued prior to the first on-study scan, and 3 subjects are currently on treatment but have not completed the first on-study imaging assessment. Bars represent the best change from baseline in the size of the target lesions using RECIST v1.1.

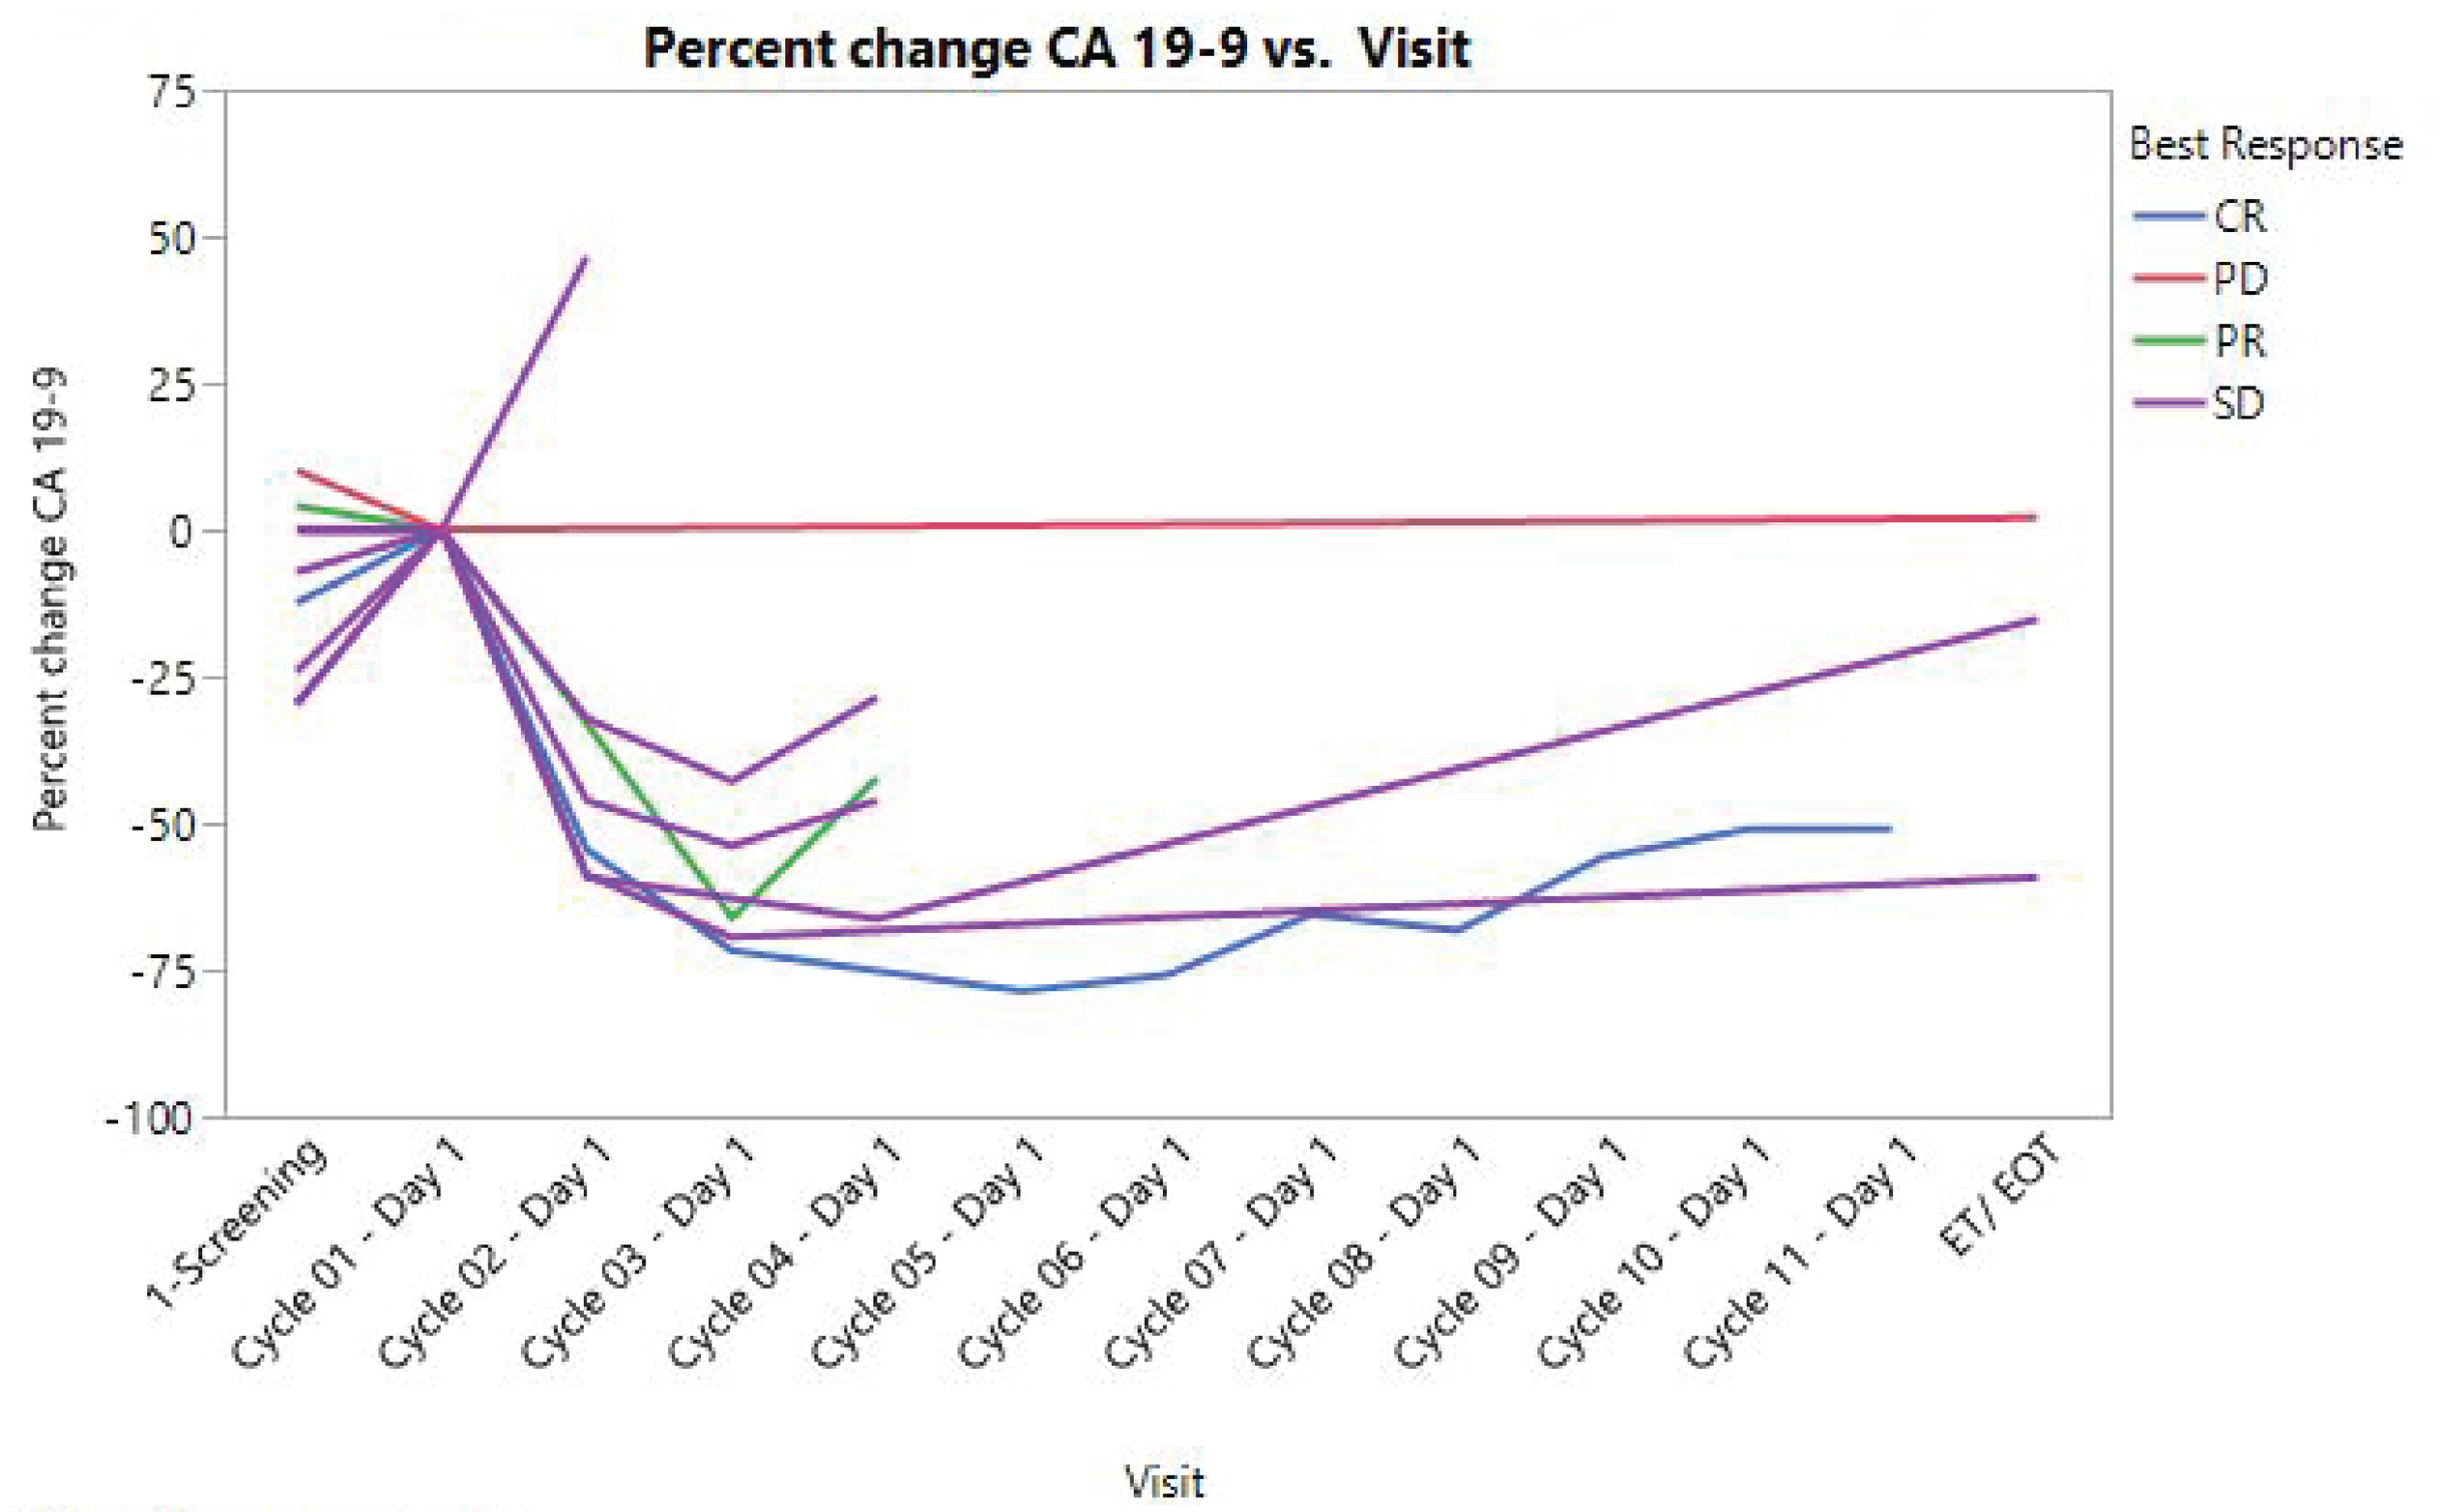
Percent change CA 19-9 vs. Visit
Percent change CA 19-9
75
50
25
0
-25
-50
-75
-100
1-Screening
Cycle 01 - Day 1
Cycle 02 - Day 1
Cycle 03 - Day 1
Cycle 04 - Day 1
Cycle 05 - Day 1
Cycle 06 - Day 1
Cycle 07 - Day 1
Cycle 08 - Day 1
Cycle 09 - Day 1
Cycle 10 - Day 1
Cycle 11 - Day 1
ET/ EOT
Visit
Best Response
CR
PD
PR
SD

CR: Complete Response, ET/EOT: End of Treatment or End of Trial Visit; PD: Progressive Disease, PR: Partial Response, SD: Stable Disease

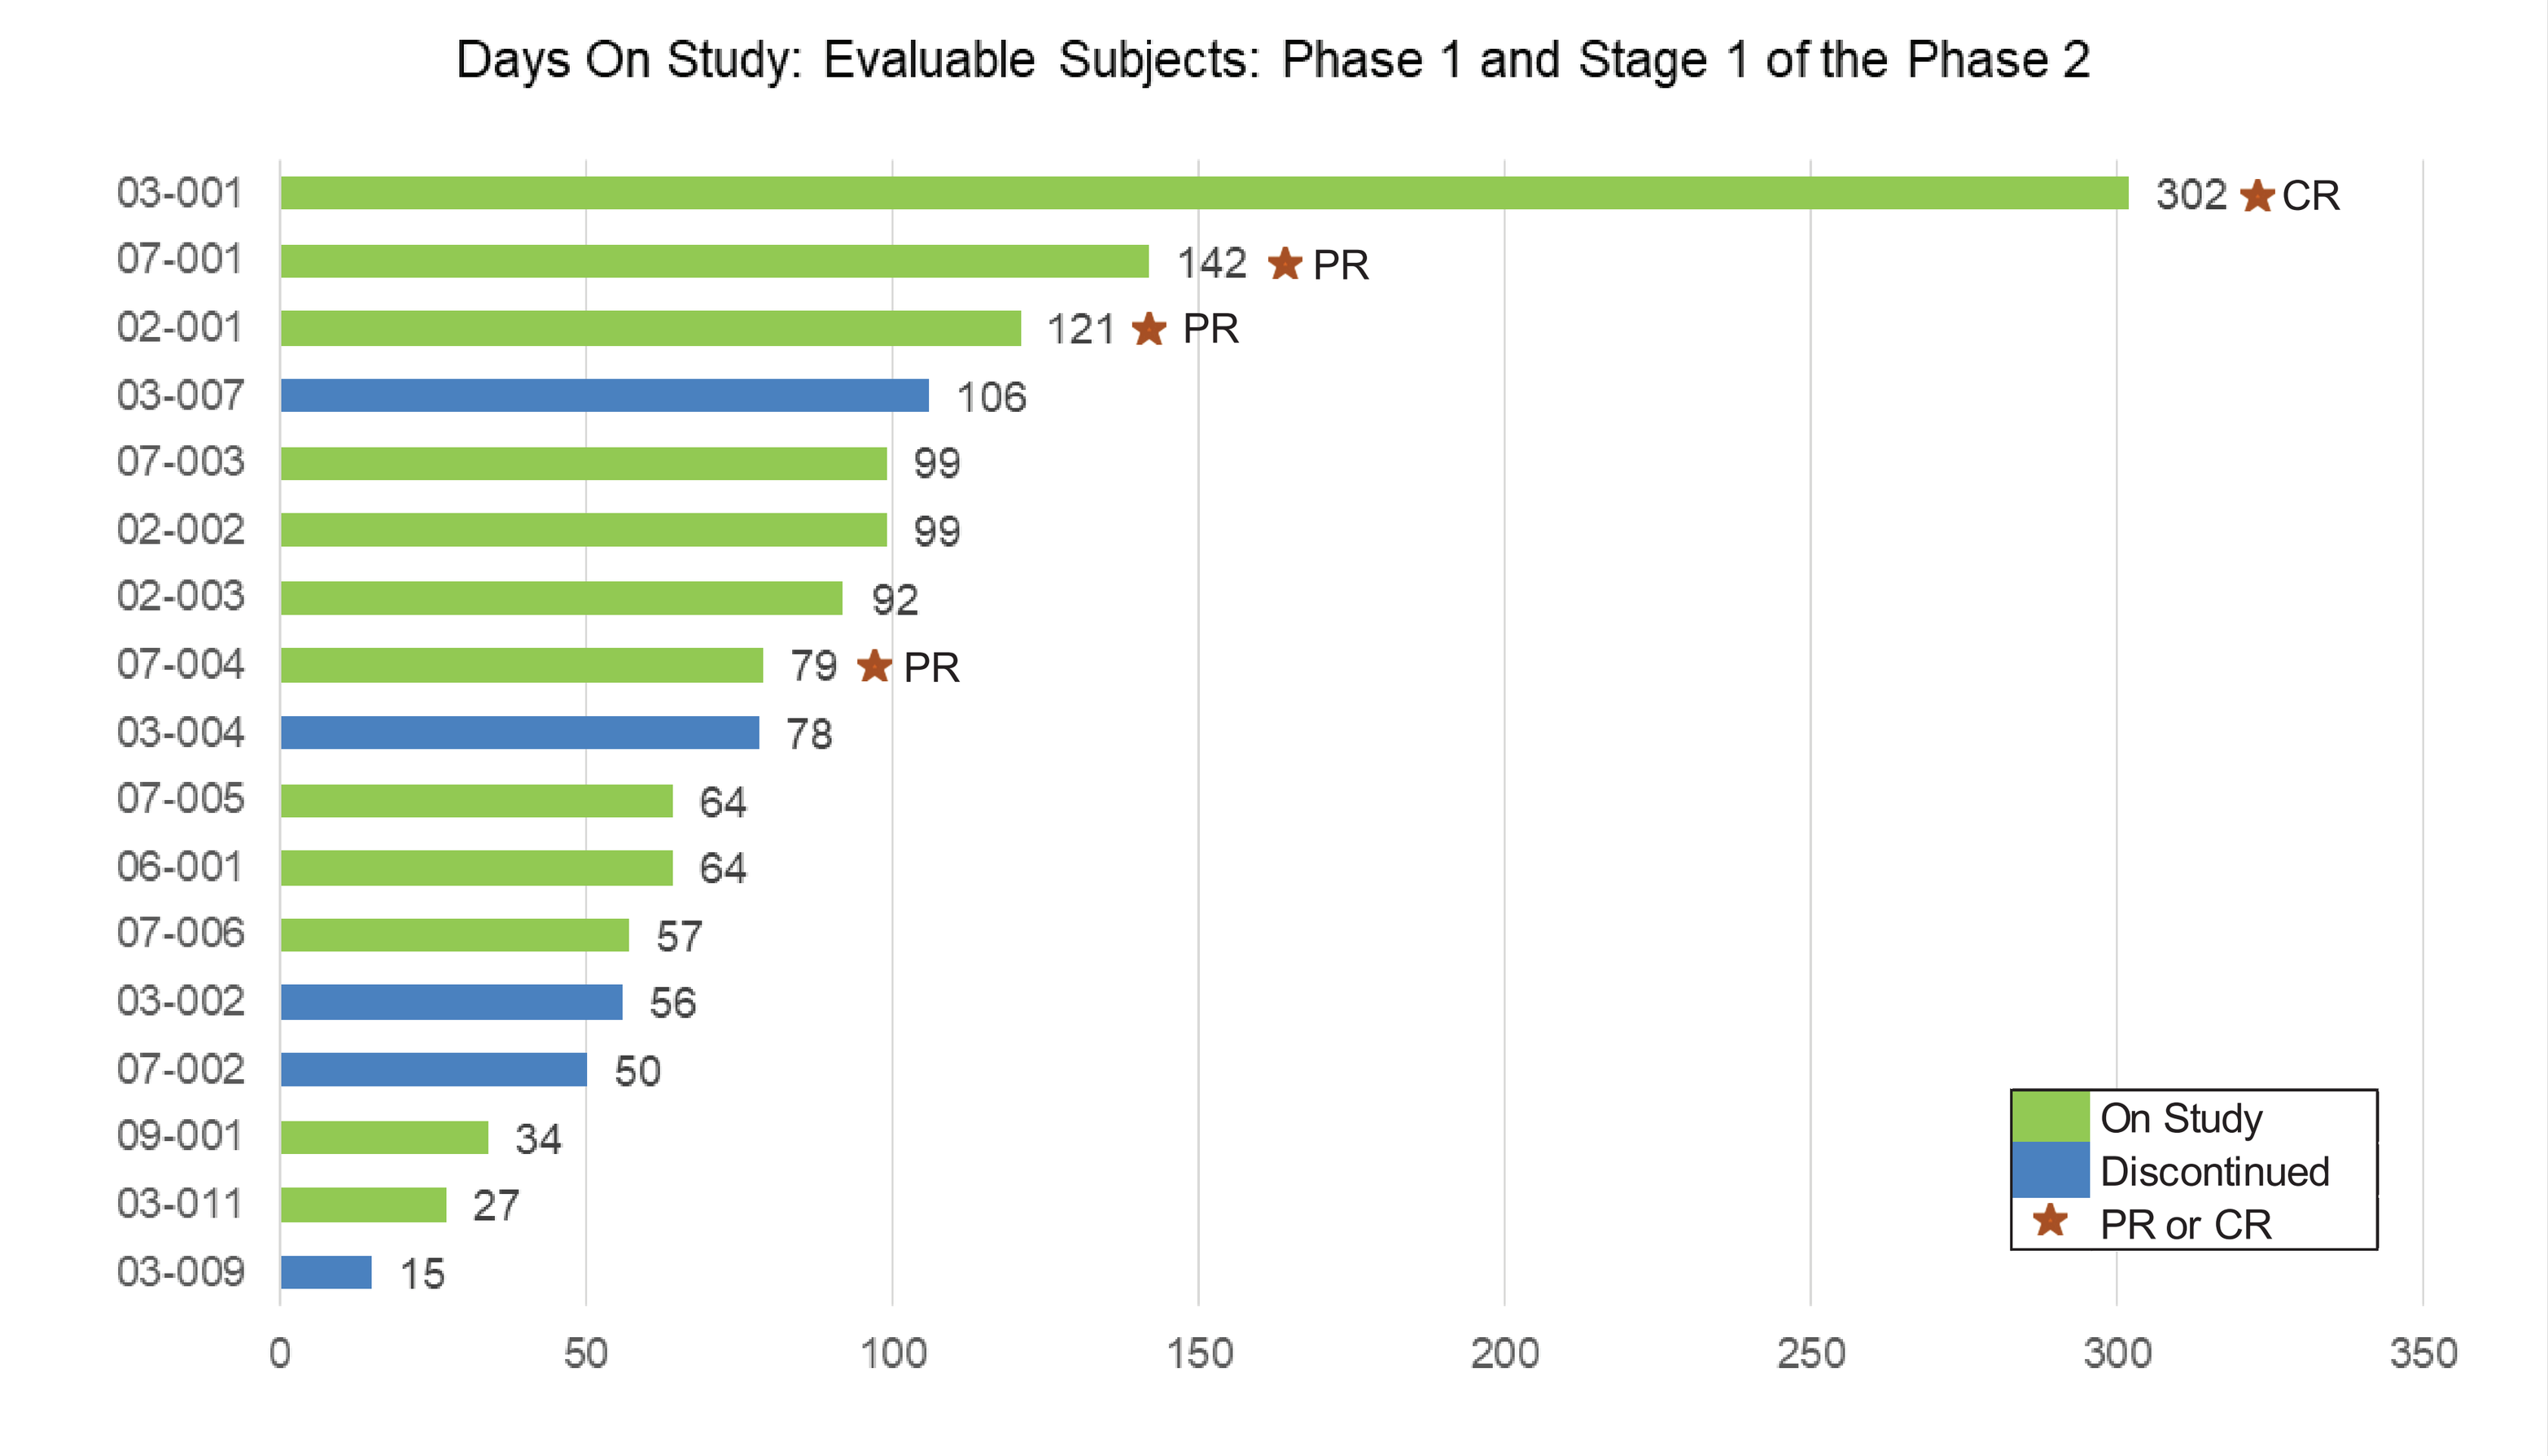
Days On Study: Evaluable Subjects: Phase 1 and Stage 1 of the Phase 2
03-001 302 CR
07-001 142 PR
02-001 121 PR
03-007 106
07-003 99
02-002 99
02-003 92
07-004 79 PR
03-004 78
07-005 64
06-001 64
07-006 57
03-002 56
07-002 50
09-001 34
03-011 27
03-009 15
0
50
100
150
200
250
300
350
On Study
Discontinued
PR or CR

- As of 19 September 2018, in the response evaluable population (14 subjects) from the Phase 1 and Stage 1 of the Phase 2:
 - Overall response rate was 29%
 - 1 CR was observed after 6 cycles of therapy and confirmed after 8 cycles of therapy (see Fig. A and B below)
 - 2 PRs were observed after 2 cycles of therapy, 1 PR was observed after 4 cycles of therapy
 - Disease control rate was 86% (1 Complete Response/ 3 Partial Response/ 8 Stable Disease)
 - 12 subjects had stable disease or better for at least 2 cycles (2 months)
 - Most subjects with CA 19-9 results exhibited reductions after 1 cycle of therapy (-32% to -59%). One subject experienced a 46% increase but had stable disease after 2 cycles.

Conclusions

- The combination of RX-3117 and nab-pac appears safe and well tolerated when administered at the recommended Phase 2 dose of RX-3117: 700 mg for 5 consecutive days with 2 days off for 4 weeks and nab-pac at 125mg/m^2 in each 4 week cycle in combination with nab-Pac
- The Phase 2- Stage 1 endpoint was met since 2 (out of 7) subjects had partial responses thus meeting the protocol defined response criteria..
- The study continues to enroll patients with metastatic pancreatic cancer in Stage 2.

Author Disclosures: Ely Benaim, MD, Callie Heaton, and Christine Peterson, PhD – Rexahn Pharmaceuticals

For further information about RX-3117 and Rexahn Pharmaceuticals
please contact Dr. Ely Benaim: benaime@rexahn.com, (240) 268-5300 x 304