Medscape EDUCATION

C difficile Prevention and Management: An Assessment of Current Clinical Practice Patterns of Physicians

LUBARDA JOVANA[1], MAEGLIN JOHN[1], MATHEWS DEBRA[2], DELK TRUDI[2], SEPTIMUS EDWARD[3]

[1]Medscape LLC, New York, United States; [2]Synthetic Biologics, Maryland, United States;
[3]Texas A&M College of Medicine, Internal Medicine, Houston, United States

BACKGROUND & AIMS

The aim of this study was to investigate physicians' current practice patterns, knowledge, and competence in prevention and management of *Clostridium difficile* infection (CDI)

METHODS

- A clinical practice assessment consisting of 25 multiple-choice knowledge- and case-based questions was made available to US physicians in multiple specialties, including infectious disease, emergency medicine, surgery, and gastroenterology, who encounter patients with CDI. There was no monetary compensation or charge for participation
- Questions evaluated knowledge, competence, skills, barriers, and attitudes related to CDI, such as recognition of risk factors, strategies for limiting risk, and emerging strategies for prevention
- The assessment launched online on a website dedicated to continuous professional development on October 27, 2017. Data were collected until January 16, 2018
- Respondent confidentiality was maintained and responses were de-identified and aggregated prior to analyses

N = 1115 PHYSICIANS
(infectious disease, emergency medicine, surgery and gastroenterology)

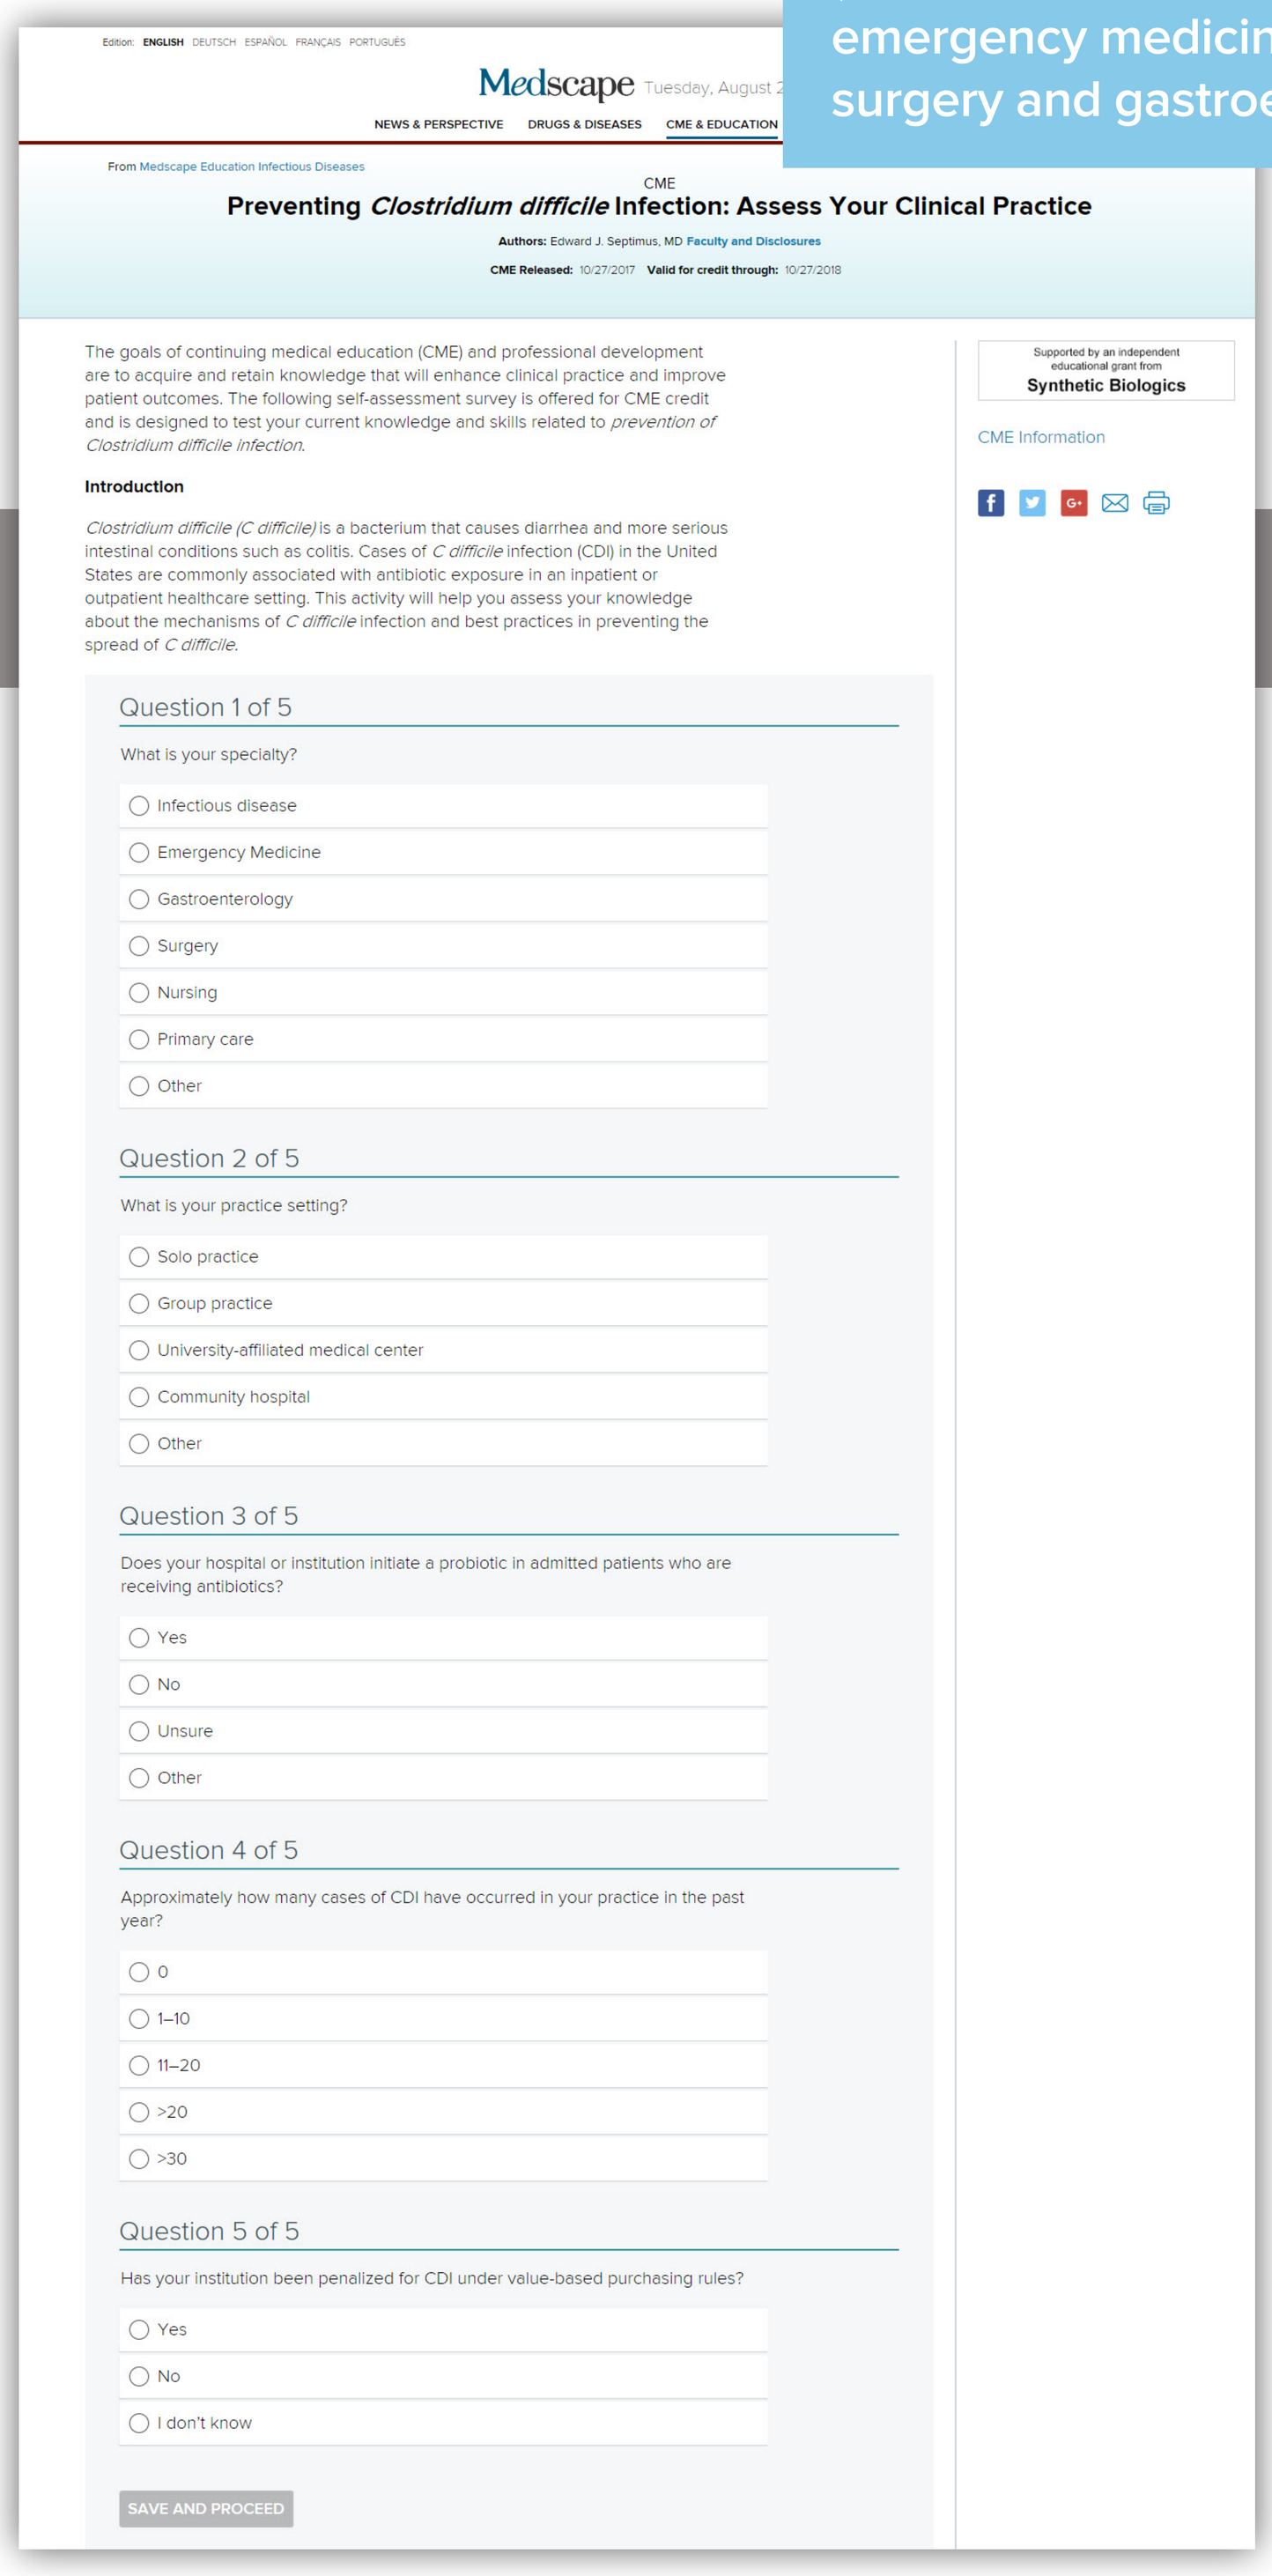

Medscape
CME
Preventing *Clostridium difficile* Infection: Assess Your Clinical Practice
Introduction
Question 1 of 5
What is your specialty?
Question 2 of 5
What is your practice setting?
Question 3 of 5
Question 4 of 5
Approximately how many cases of CDI have occurred in your practice in the past year?
Question 5 of 5
Has your institution been penalized for CDI under value-based purchasing rules?
SAVE AND PROCEED

RESULTS

INCIDENCE OF CDI

69% were not aware of the incidence of CDI in the United States

Which of the following is an accurate statement about CDI in the United States?

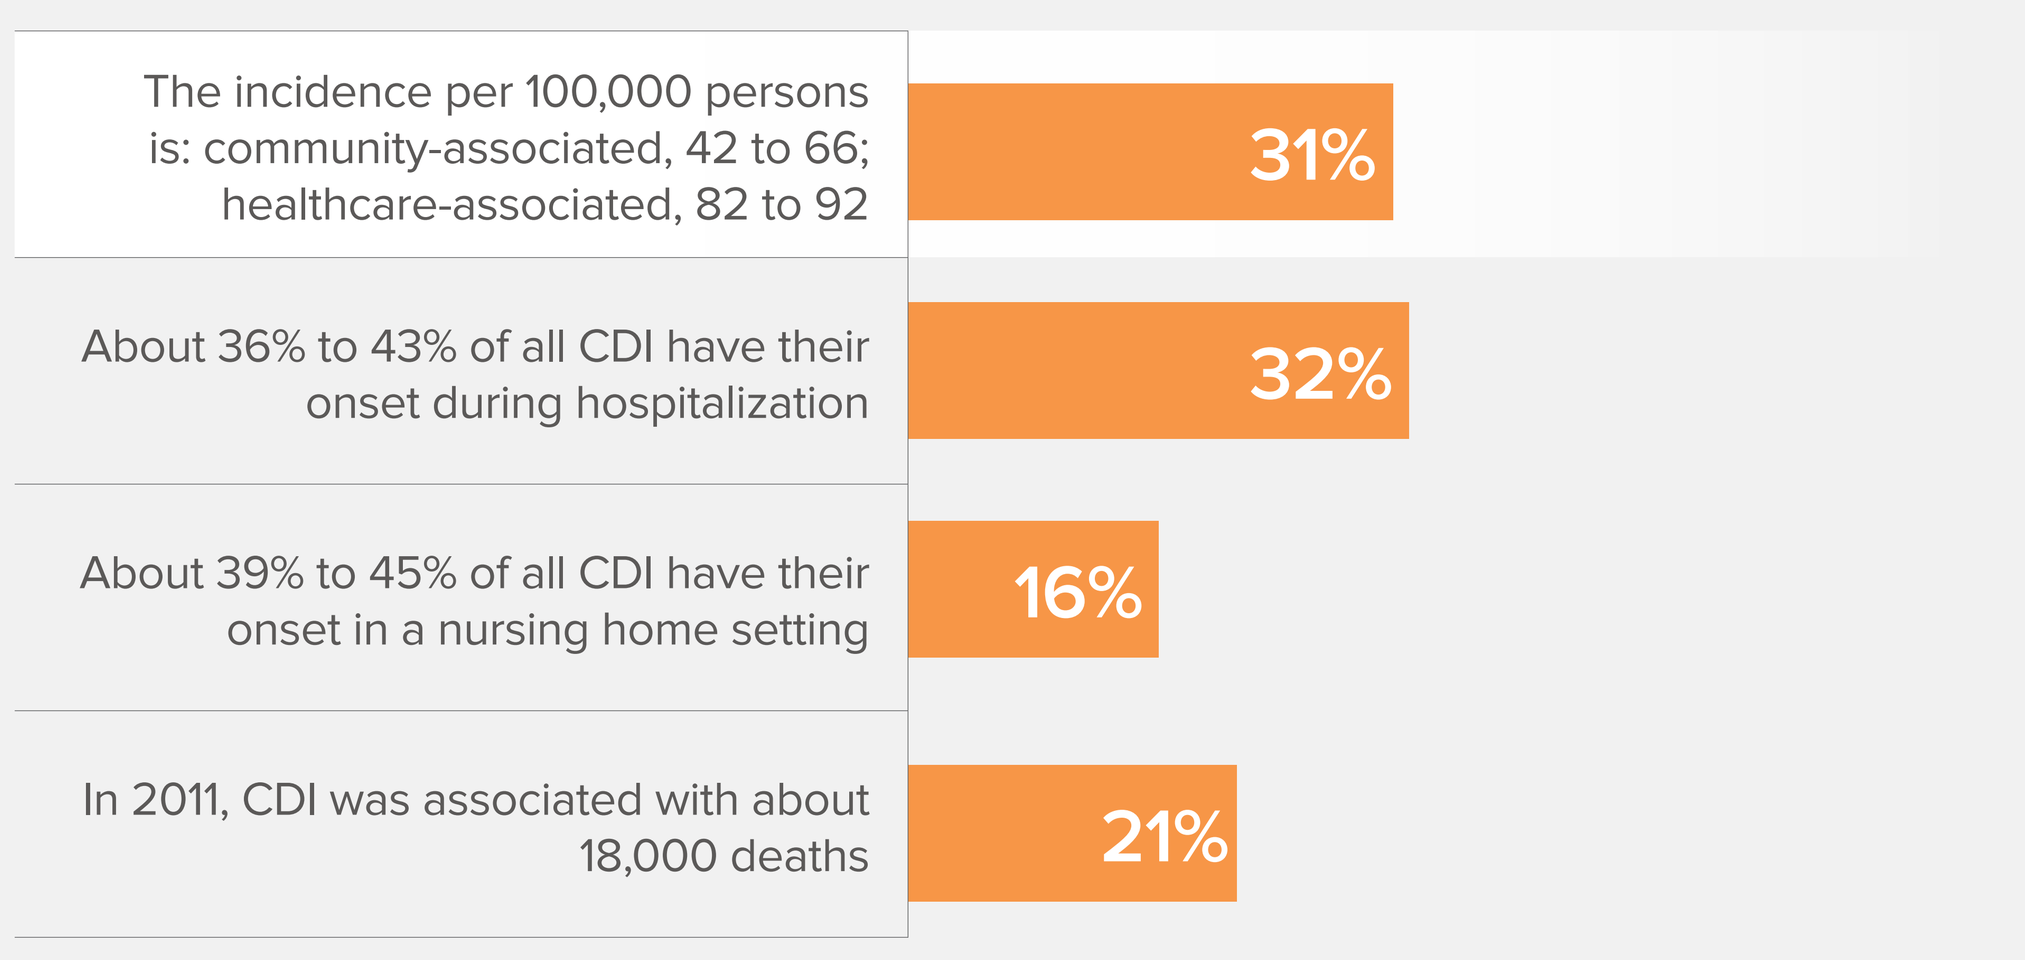

The incidence per 100,000 persons is: community-associated, 42 to 66; healthcare-associated, 82 to 92
31%
About 36% to 43% of all CDI have their onset during hospitalization
32%
About 39% to 45% of all CDI have their onset in a nursing home setting
16%
In 2011, CDI was associated with about 18,000 deaths
21%

73% reported at least 1 case of CDI occurring in their practice over the past year

Approximately how many cases of CDI have occurred in your practice in the past year?

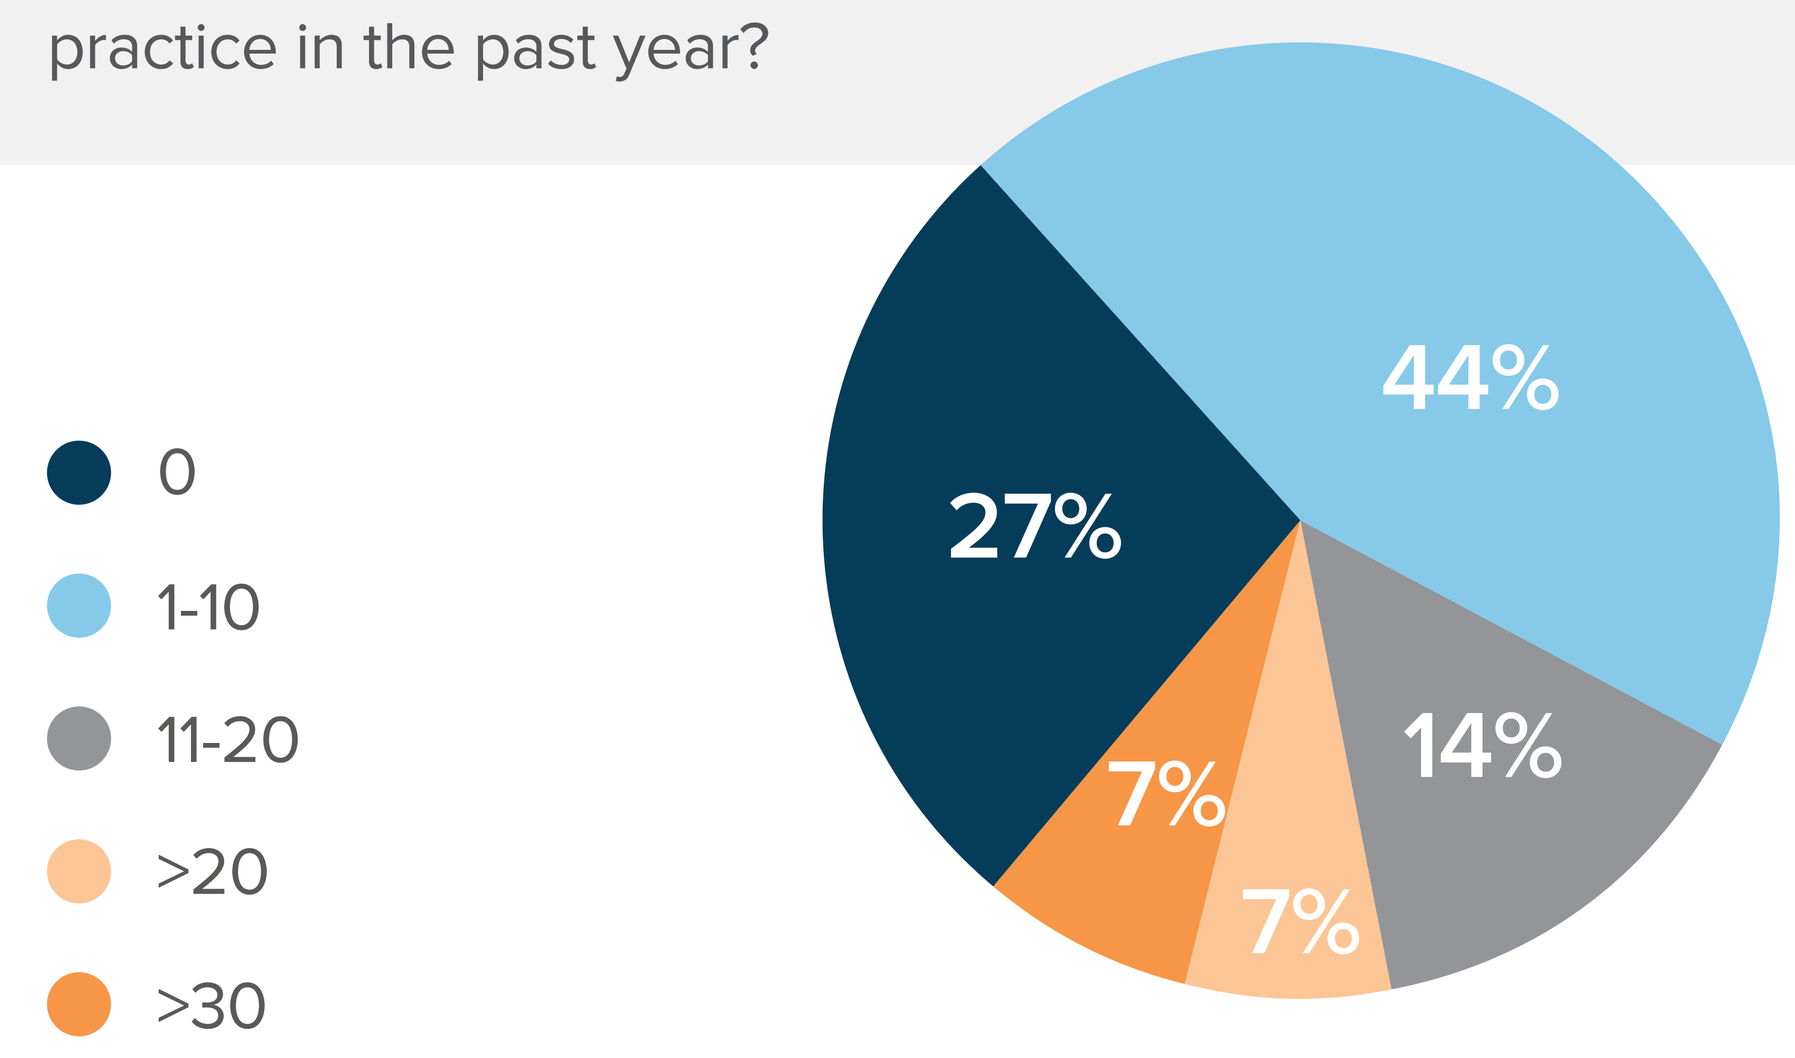

0
1-10
11-20
>20
>30
27%
44%
14%
7%
7%

RISK FACTORS FOR CDI

Majority of physicians correctly identified antibiotics most closely associated with development of CDI

The use of which of the following antibiotic or class of antibiotics is most closely associated with the development of CDI?

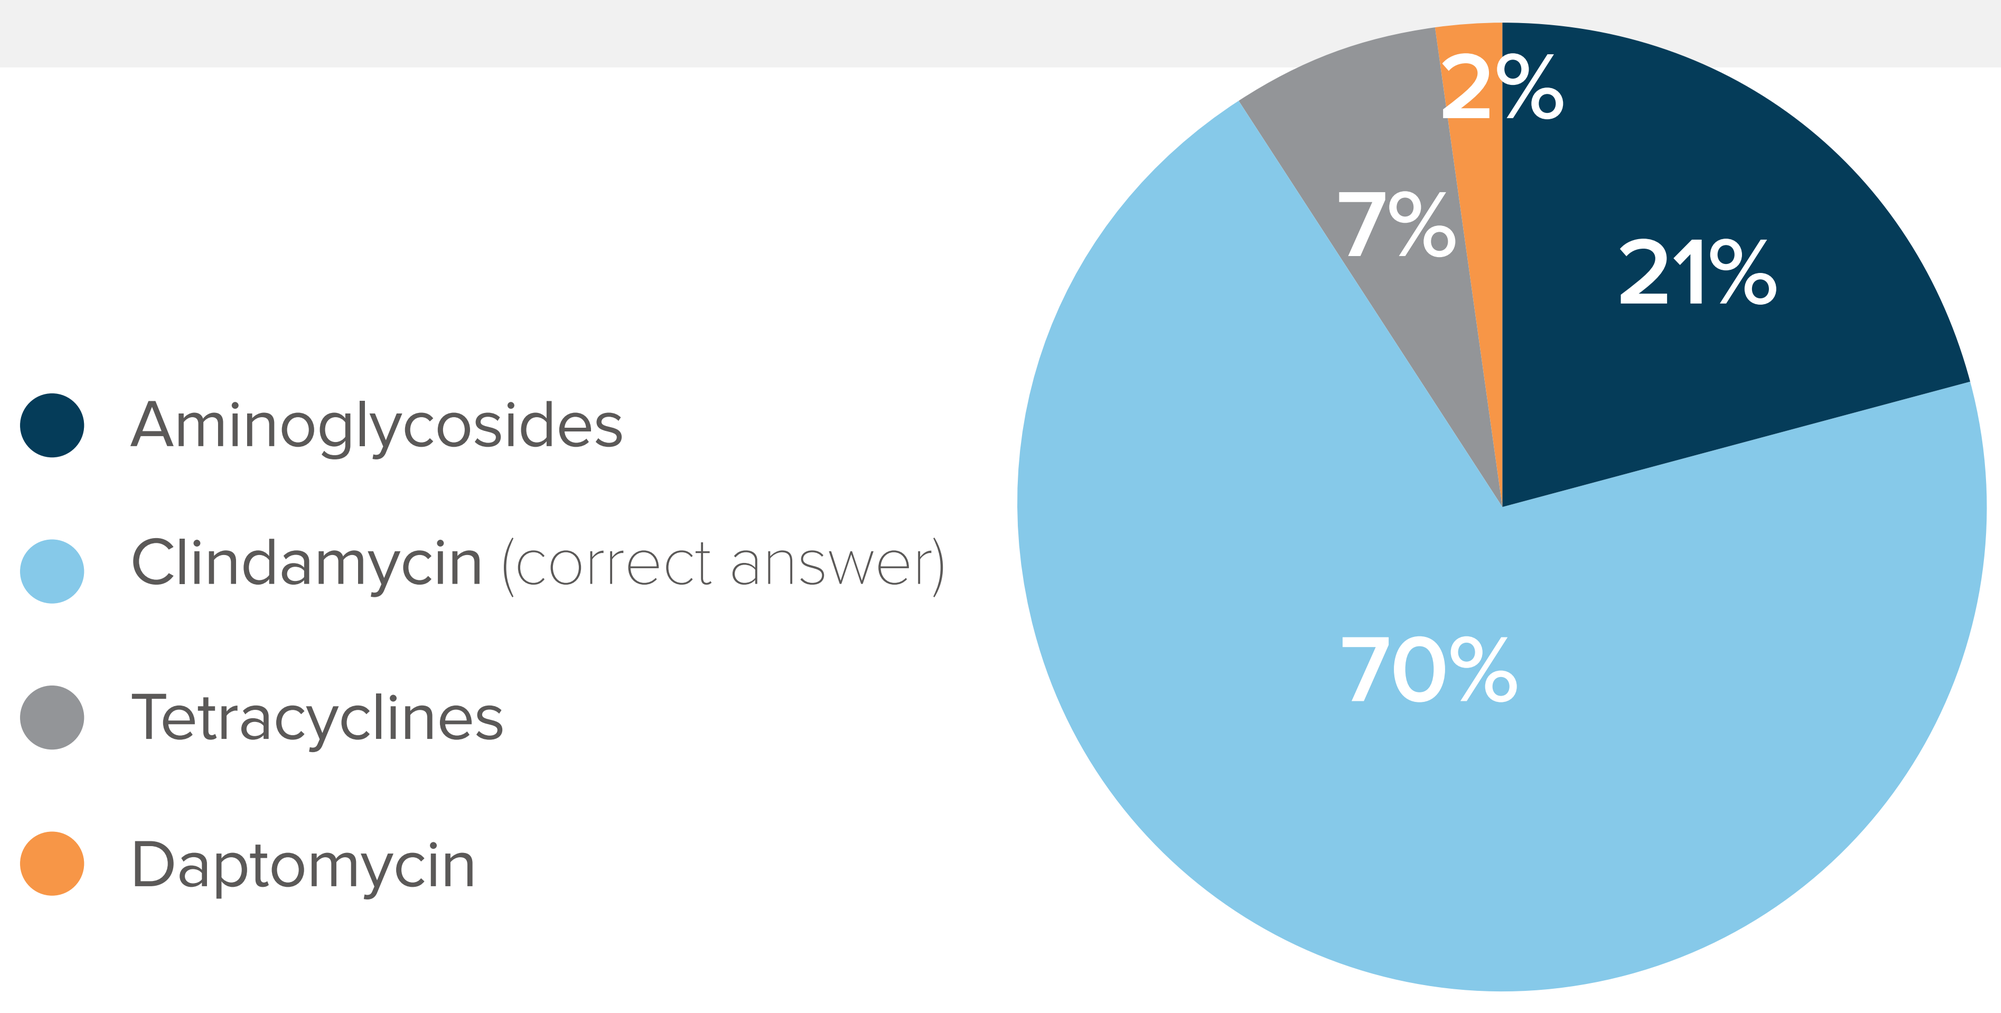

Aminoglycosides
Clindamycin (correct answer)
Tetracyclines
Daptomycin
2%
7%
21%
70%

Only 8% reported they were very confident in recognizing host risk factors for CDI

Two events are required for the development of CDI: Disruption of the fecal microbiota (typically via use of antibiotics) and ingestion of spores via the fecal-oral route. Host factors also have a role in a person's risk for CDI. How confident are you in recognizing host risk factors for CDI?

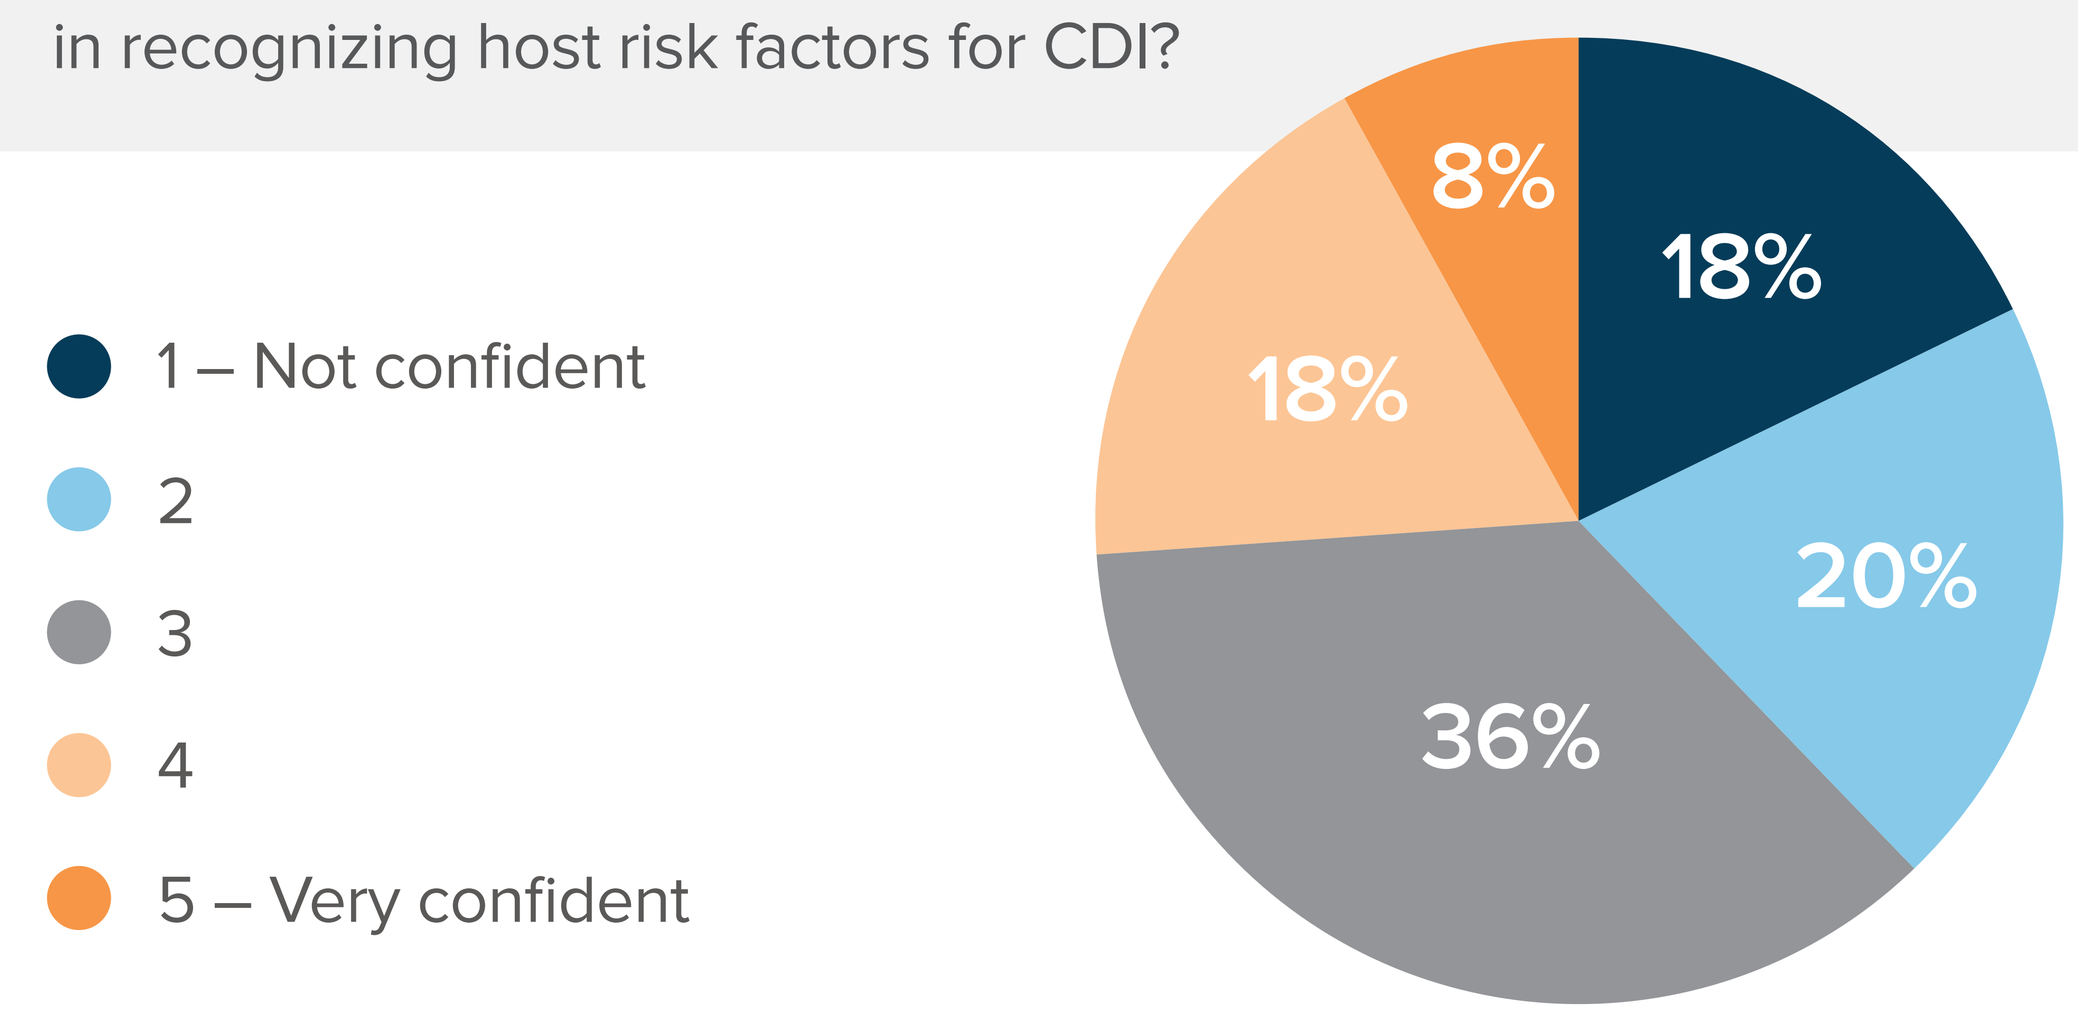

1 – Not confident
2
3
4
5 – Very confident
8%
18%
18%
20%
36%

57% were not aware of the relationship between the gut microbiome and CDI

Which of the following is not an accurate statement about the relationship between gut microbiota and CDI?

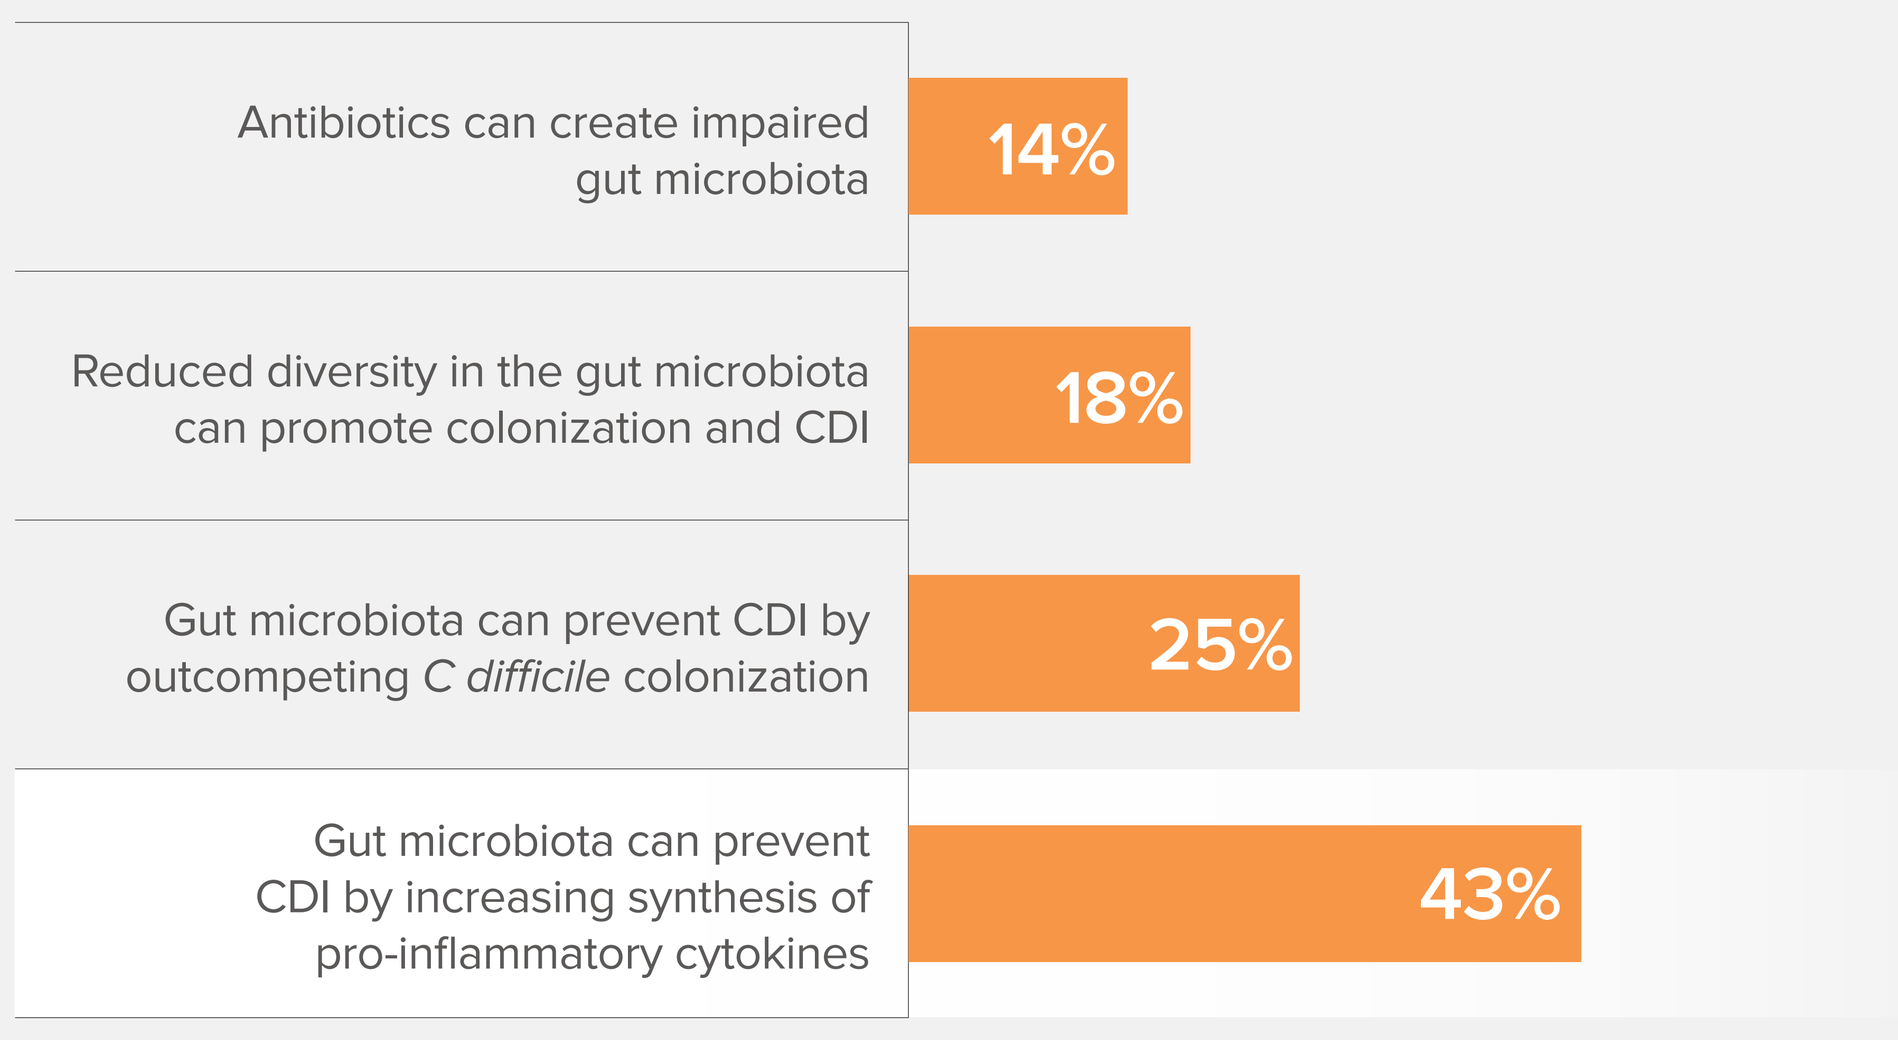

Antibiotics can create impaired gut microbiota
14%
Reduced diversity in the gut microbiota can promote colonization and CDI
18%
Gut microbiota can prevent CDI by outcompeting *C difficile* colonization
25%
Gut microbiota can prevent CDI by increasing synthesis of pro-inflammatory cytokines
43%

DIAGNOSIS OF CDI

43% use a polymerase chain reaction-based method for CDI diagnosis; 29% use a 2-step method combining different test types

What does your institution use for the diagnosis of CDI?

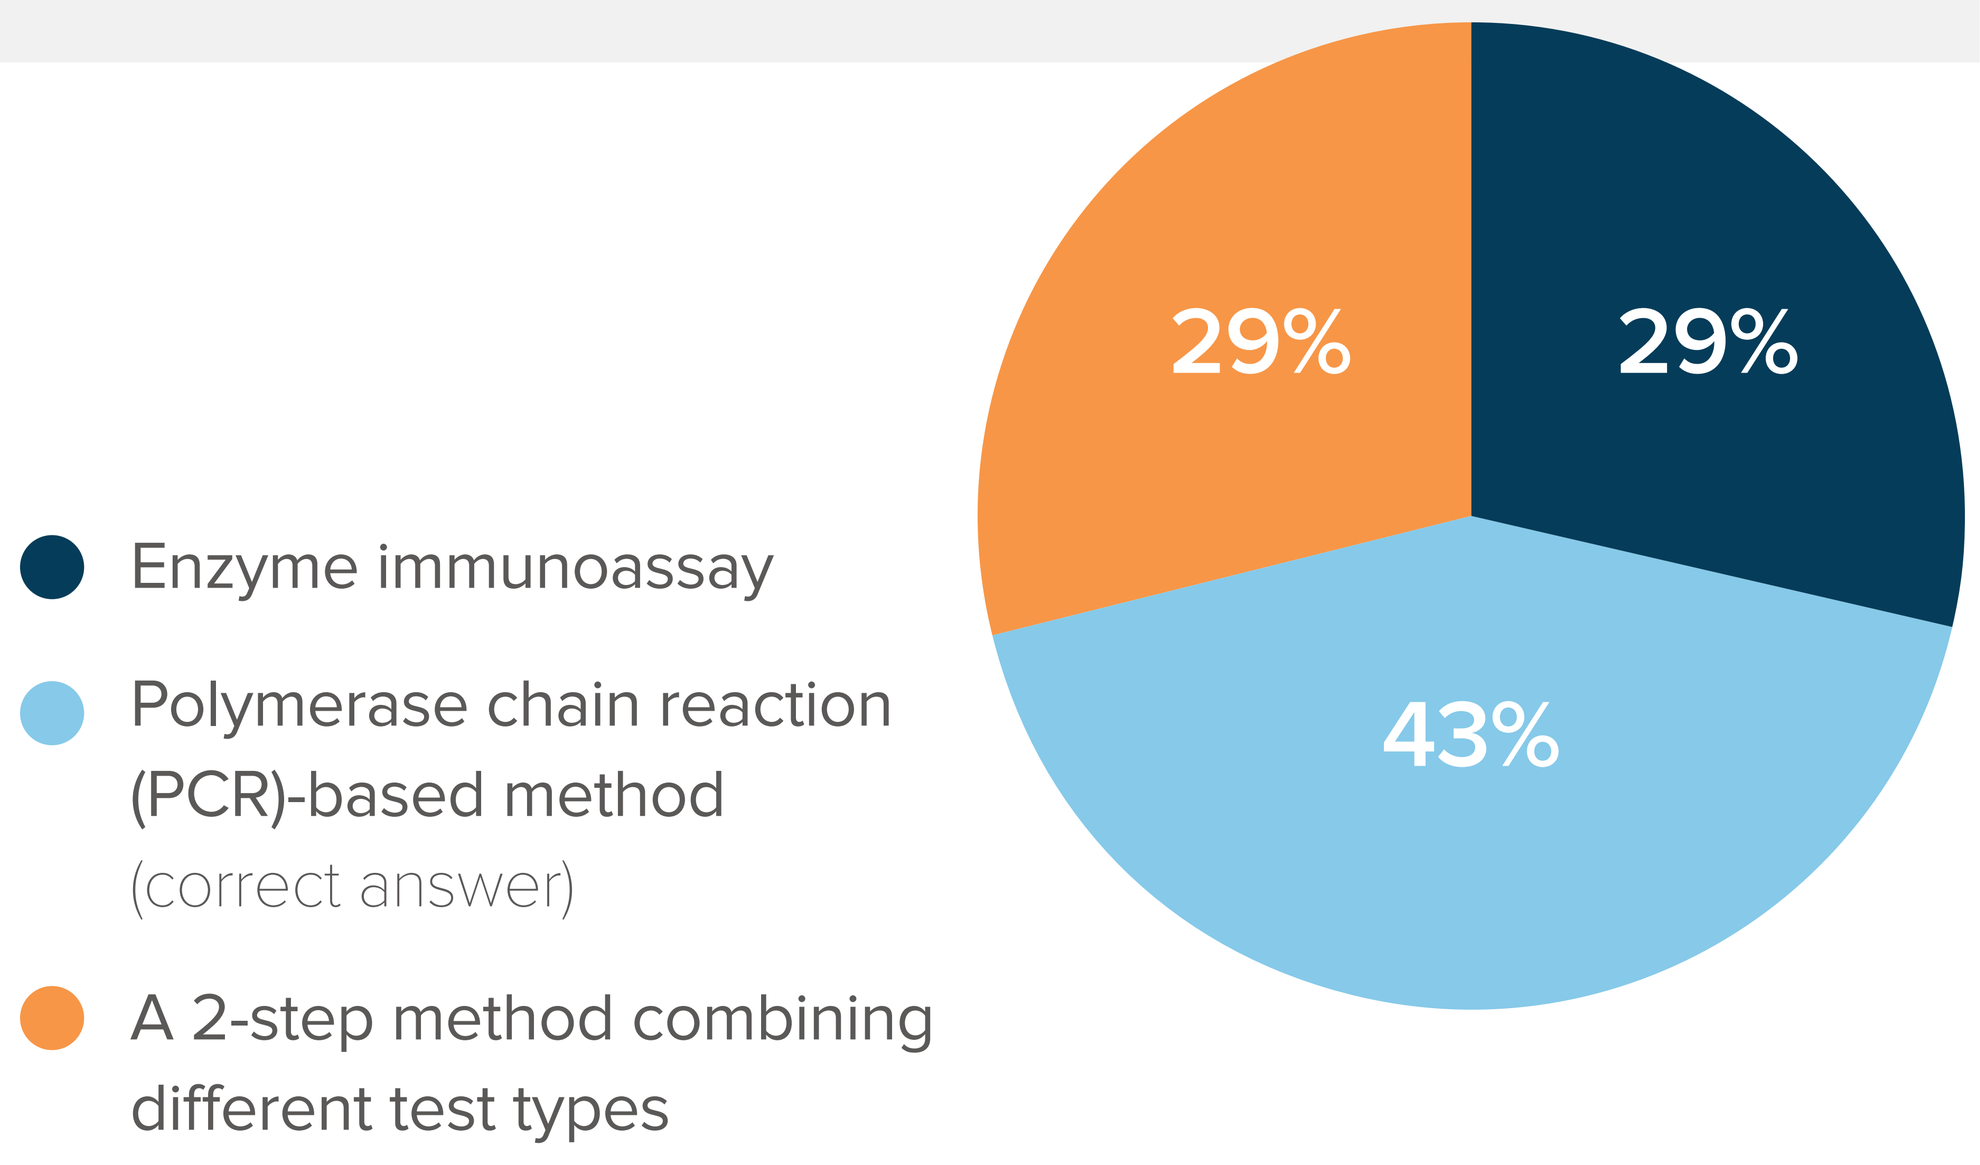

Enzyme immunoassay
Polymerase chain reaction (PCR)-based method (correct answer)
A 2-step method combining different test types
29%
29%
43%

NEW DEVELOPMENTS

58% are not aware of new strategies being investigated for prevention of CDI

Cephalosporins, penicillins, and carbapenems are beta-lactam antibiotics that are proven to damage gut microbiome diversity. Which of the following is a potential new strategy for reducing the negative effect of these medications on the gut microbiota?

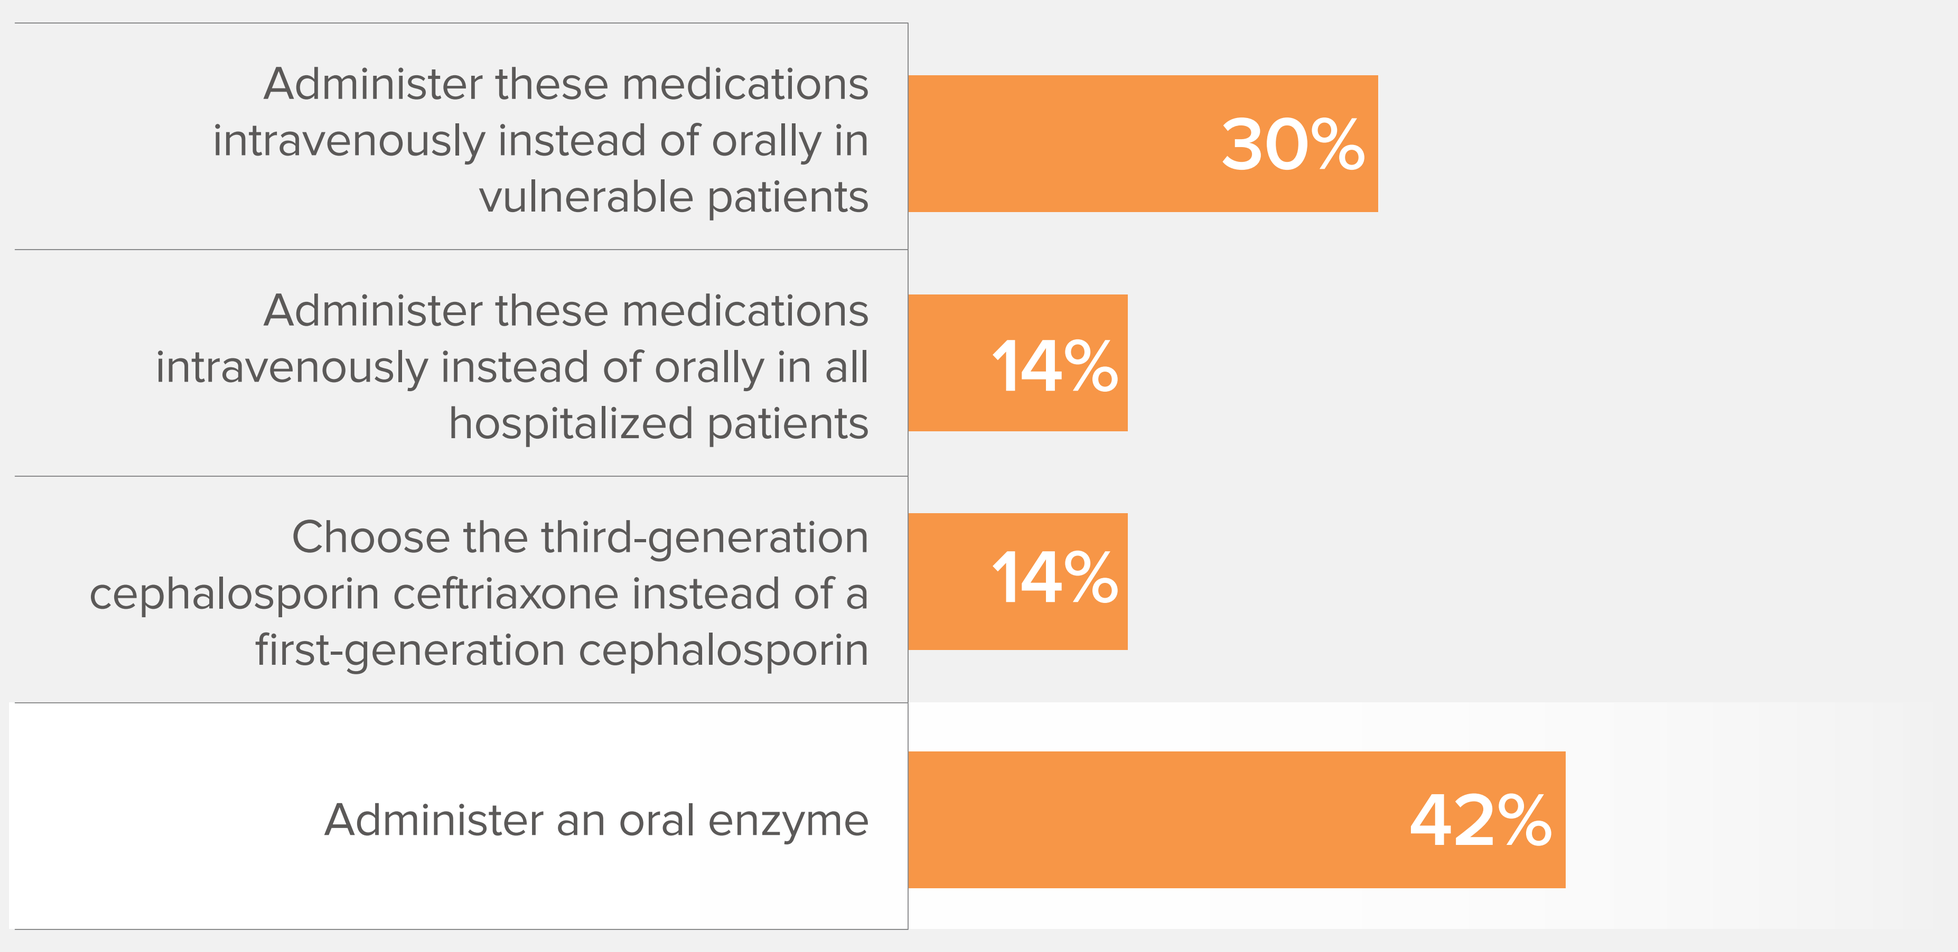

Administer these medications intravenously instead of orally in vulnerable patients
30%
Administer these medications intravenously instead of orally in all hospitalized patients
14%
Choose the third-generation cephalosporin ceftriaxone instead of a first-generation cephalosporin
14%
Administer an oral enzyme
42%

48% are not aware of the mechanism by which new therapies prevent development of CDI

What is the supposed mechanism by which ribaxamase, currently in clinical trials, may prevent CDI?

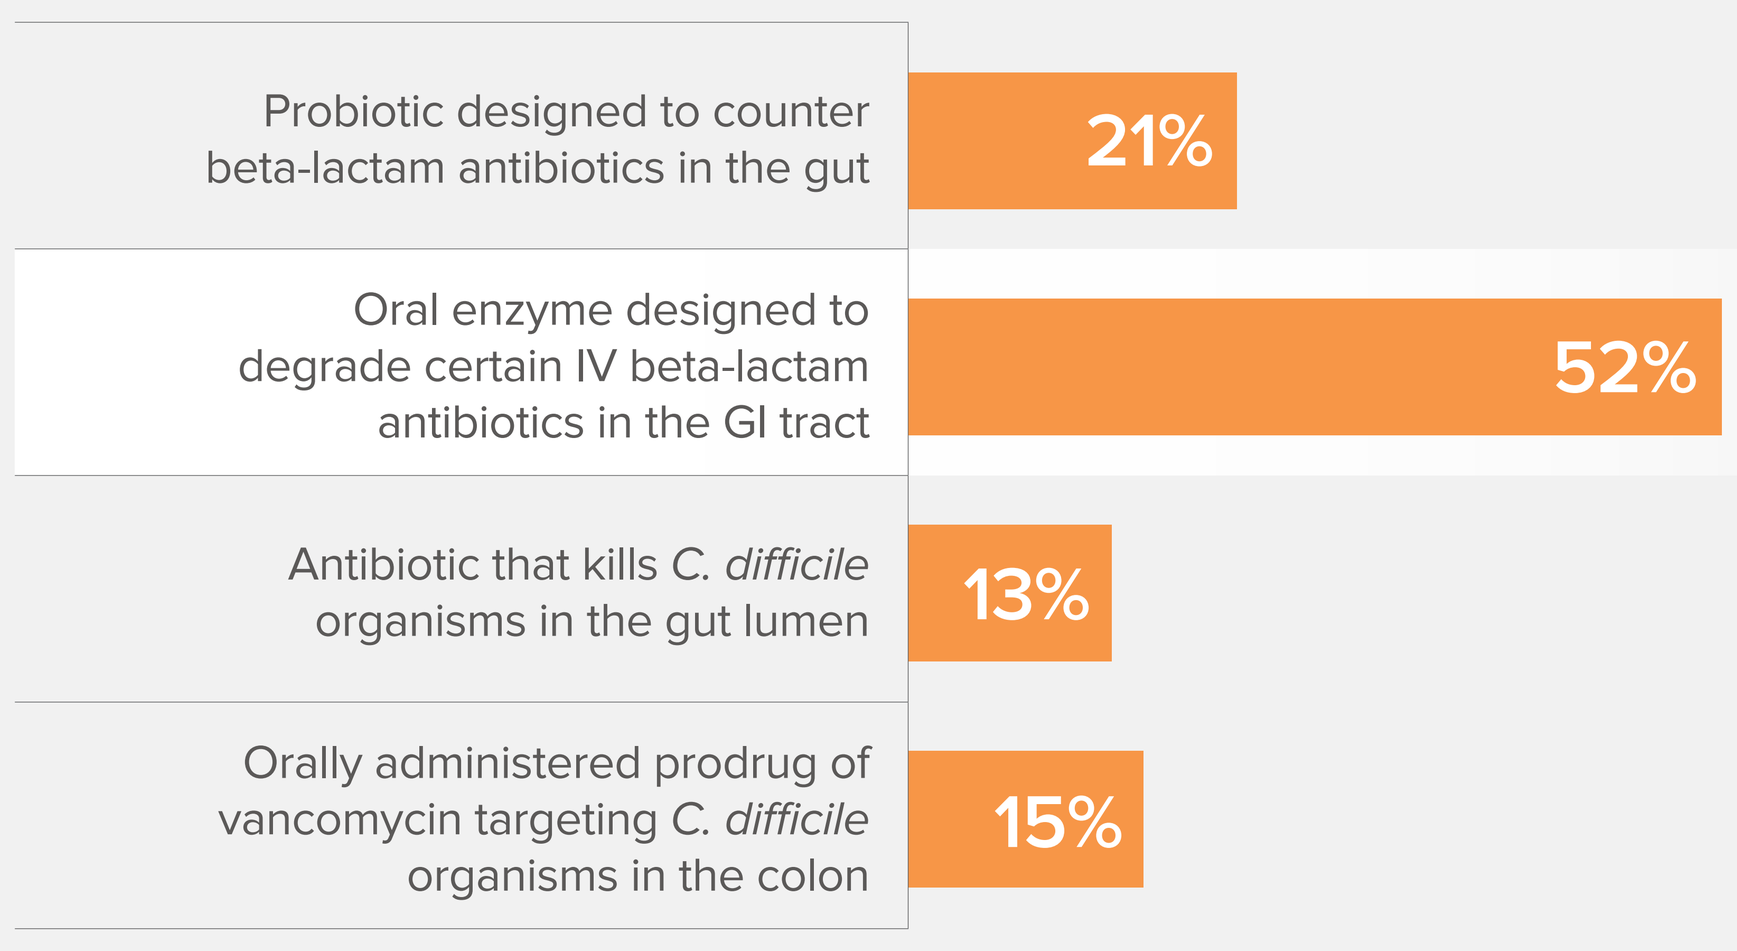

Probiotic designed to counter beta-lactam antibiotics in the gut
21%
Oral enzyme designed to degrade certain IV beta-lactam antibiotics in the GI tract
52%
Antibiotic that kills *C. difficile* organisms in the gut lumen
13%
Orally administered prodrug of vancomycin targeting *C. difficile* organisms in the colon
15%

CONCLUSION

This research yielded important insights into current clinical practices of physicians and gaps in the prevention and management of CDI that could inform development of future medical education projects

ACKNOWLEDGEMENTS

This clinical practice assessment research was funded through an independent educational grant from Synthetic Biologics, Inc.

For more information, contact Jovana Lubarda, PhD Director, Educational Strategy, Medscape, LLC, jlubarda@medscape.net

Scan here to view this poster online.