Efficacy signals, long-term exposure and safety data from a phase 1-2 study of a cell-penetrating peptide antagonist of CEBPβ, a novel target, in patients with refractory solid tumors

Elisa Fontana[1], Anja Williams[1], Gerald Falchook[2], Nehal Lakhani[3], Jeff Evans[4], Vinai Gondi[5], Fabio Iwamoto[6], Meredith McKean[7], Stefan Symeonides[8], Nicholas Butowski[9], Alistair McLaren[4], Robert Buerki[5], Jason Henry[2], Jim Rotolo[10], Gina Capiaux[10], Rob Michel[10], Stephen Kaesshaefer[10], Erol Wiegert[10], and Alice Bexon[10]

Background

- CCAAT/enhancer-binding protein β (C/EBPβ) is an oncogenic transcription factor that promotes cell proliferation, survival, and differentiation
- C/EBPβ regulates polarization toward immunosuppressive M2 phenotype in activated macrophage [6,7]
- ST101, a cell-penetrating peptide, inhibits C/EBPβ dimerization and DNA binding with oncogenic co-factors (i.e., ATF5), reducing the expression of selective oncogenes
- In vitro data demonstrates ST101 targeted killing of tumor cells, across multiple cancers, while preserving normal cells
- ST101 potently activates CD8+ T-cells leading to immune activation

Rationale for targeting C/EBPβ

- Previously undruggable target implicated in suppressing anti-tumor immune response and enhancing progression of many tumors
- C/EBPβ expression has been shown to be elevated in BC, GBM, CRPC and melanoma[1-5]

ST101 penetrates cells to reach its target

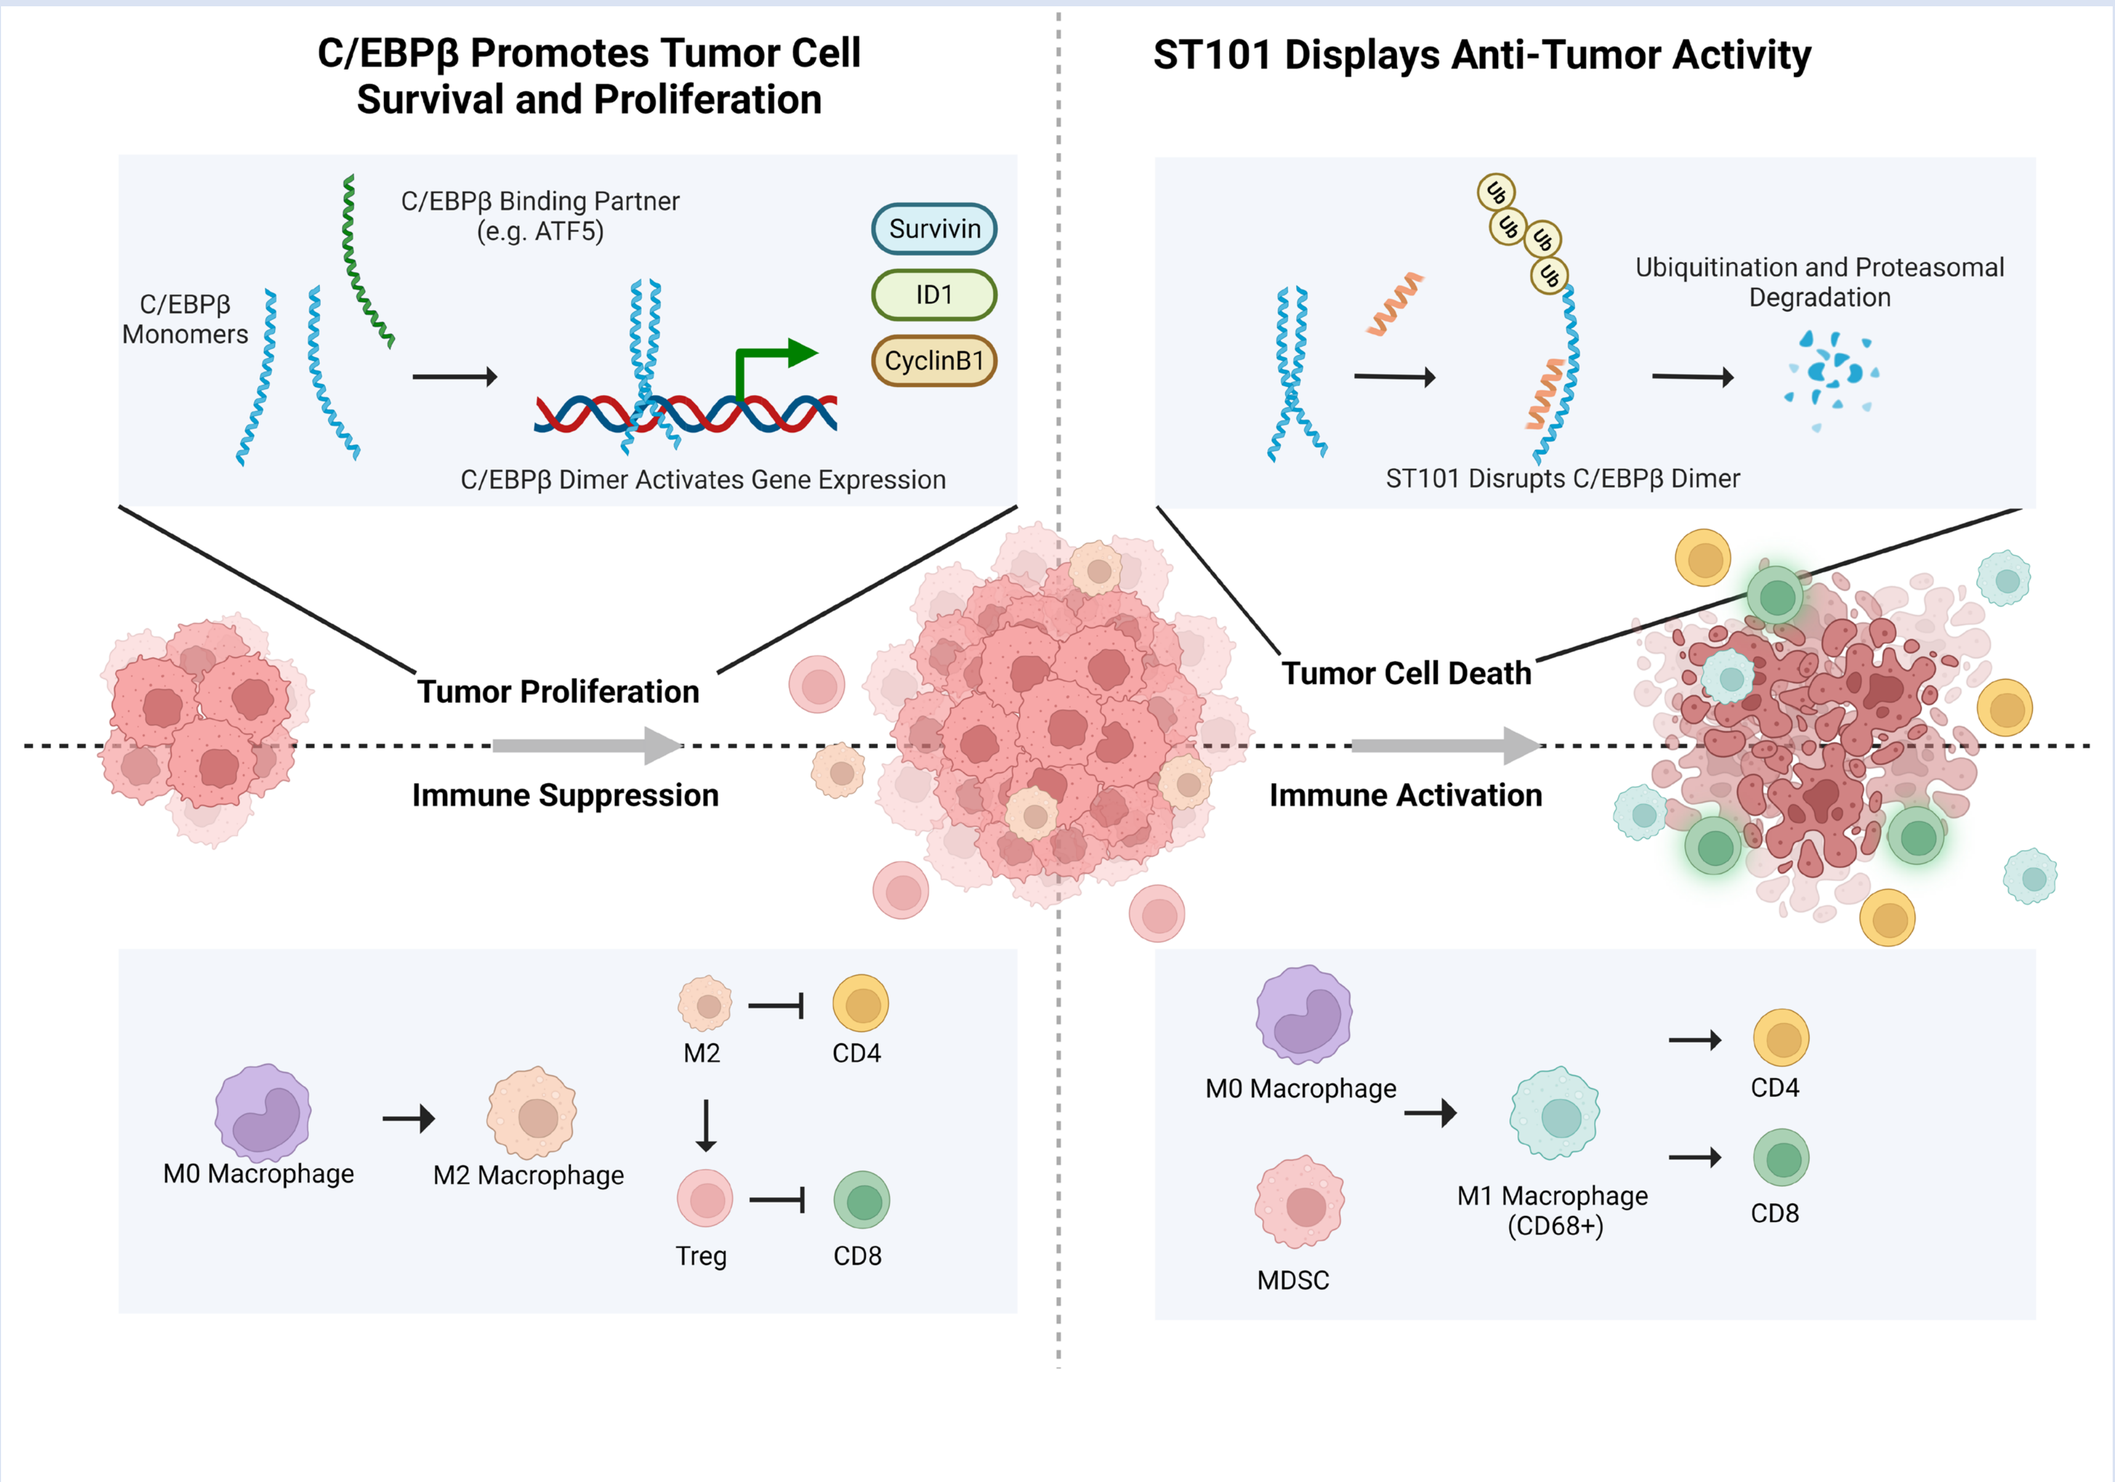
C/EBPβ Promotes Tumor Cell Survival and Proliferation
ST101 Displays Anti-Tumor Activity
C/EBPβ Binding Partner (e.g. ATF5)
C/EBPβ Monomers
Survivin
ID1
CyclinB1
C/EBPβ Dimer Activates Gene Expression
Ubiquitination and Proteasomal Degradation
ST101 Disrupts C/EBPβ Dimer
Tumor Proliferation
Immune Suppression
Tumor Cell Death
Immune Activation
M0 Macrophage
M2 Macrophage
M2
CD4
Treg
CD8
M0 Macrophage
MDSC
M1 Macrophage (CD68+)
CD4
CD8

Study design

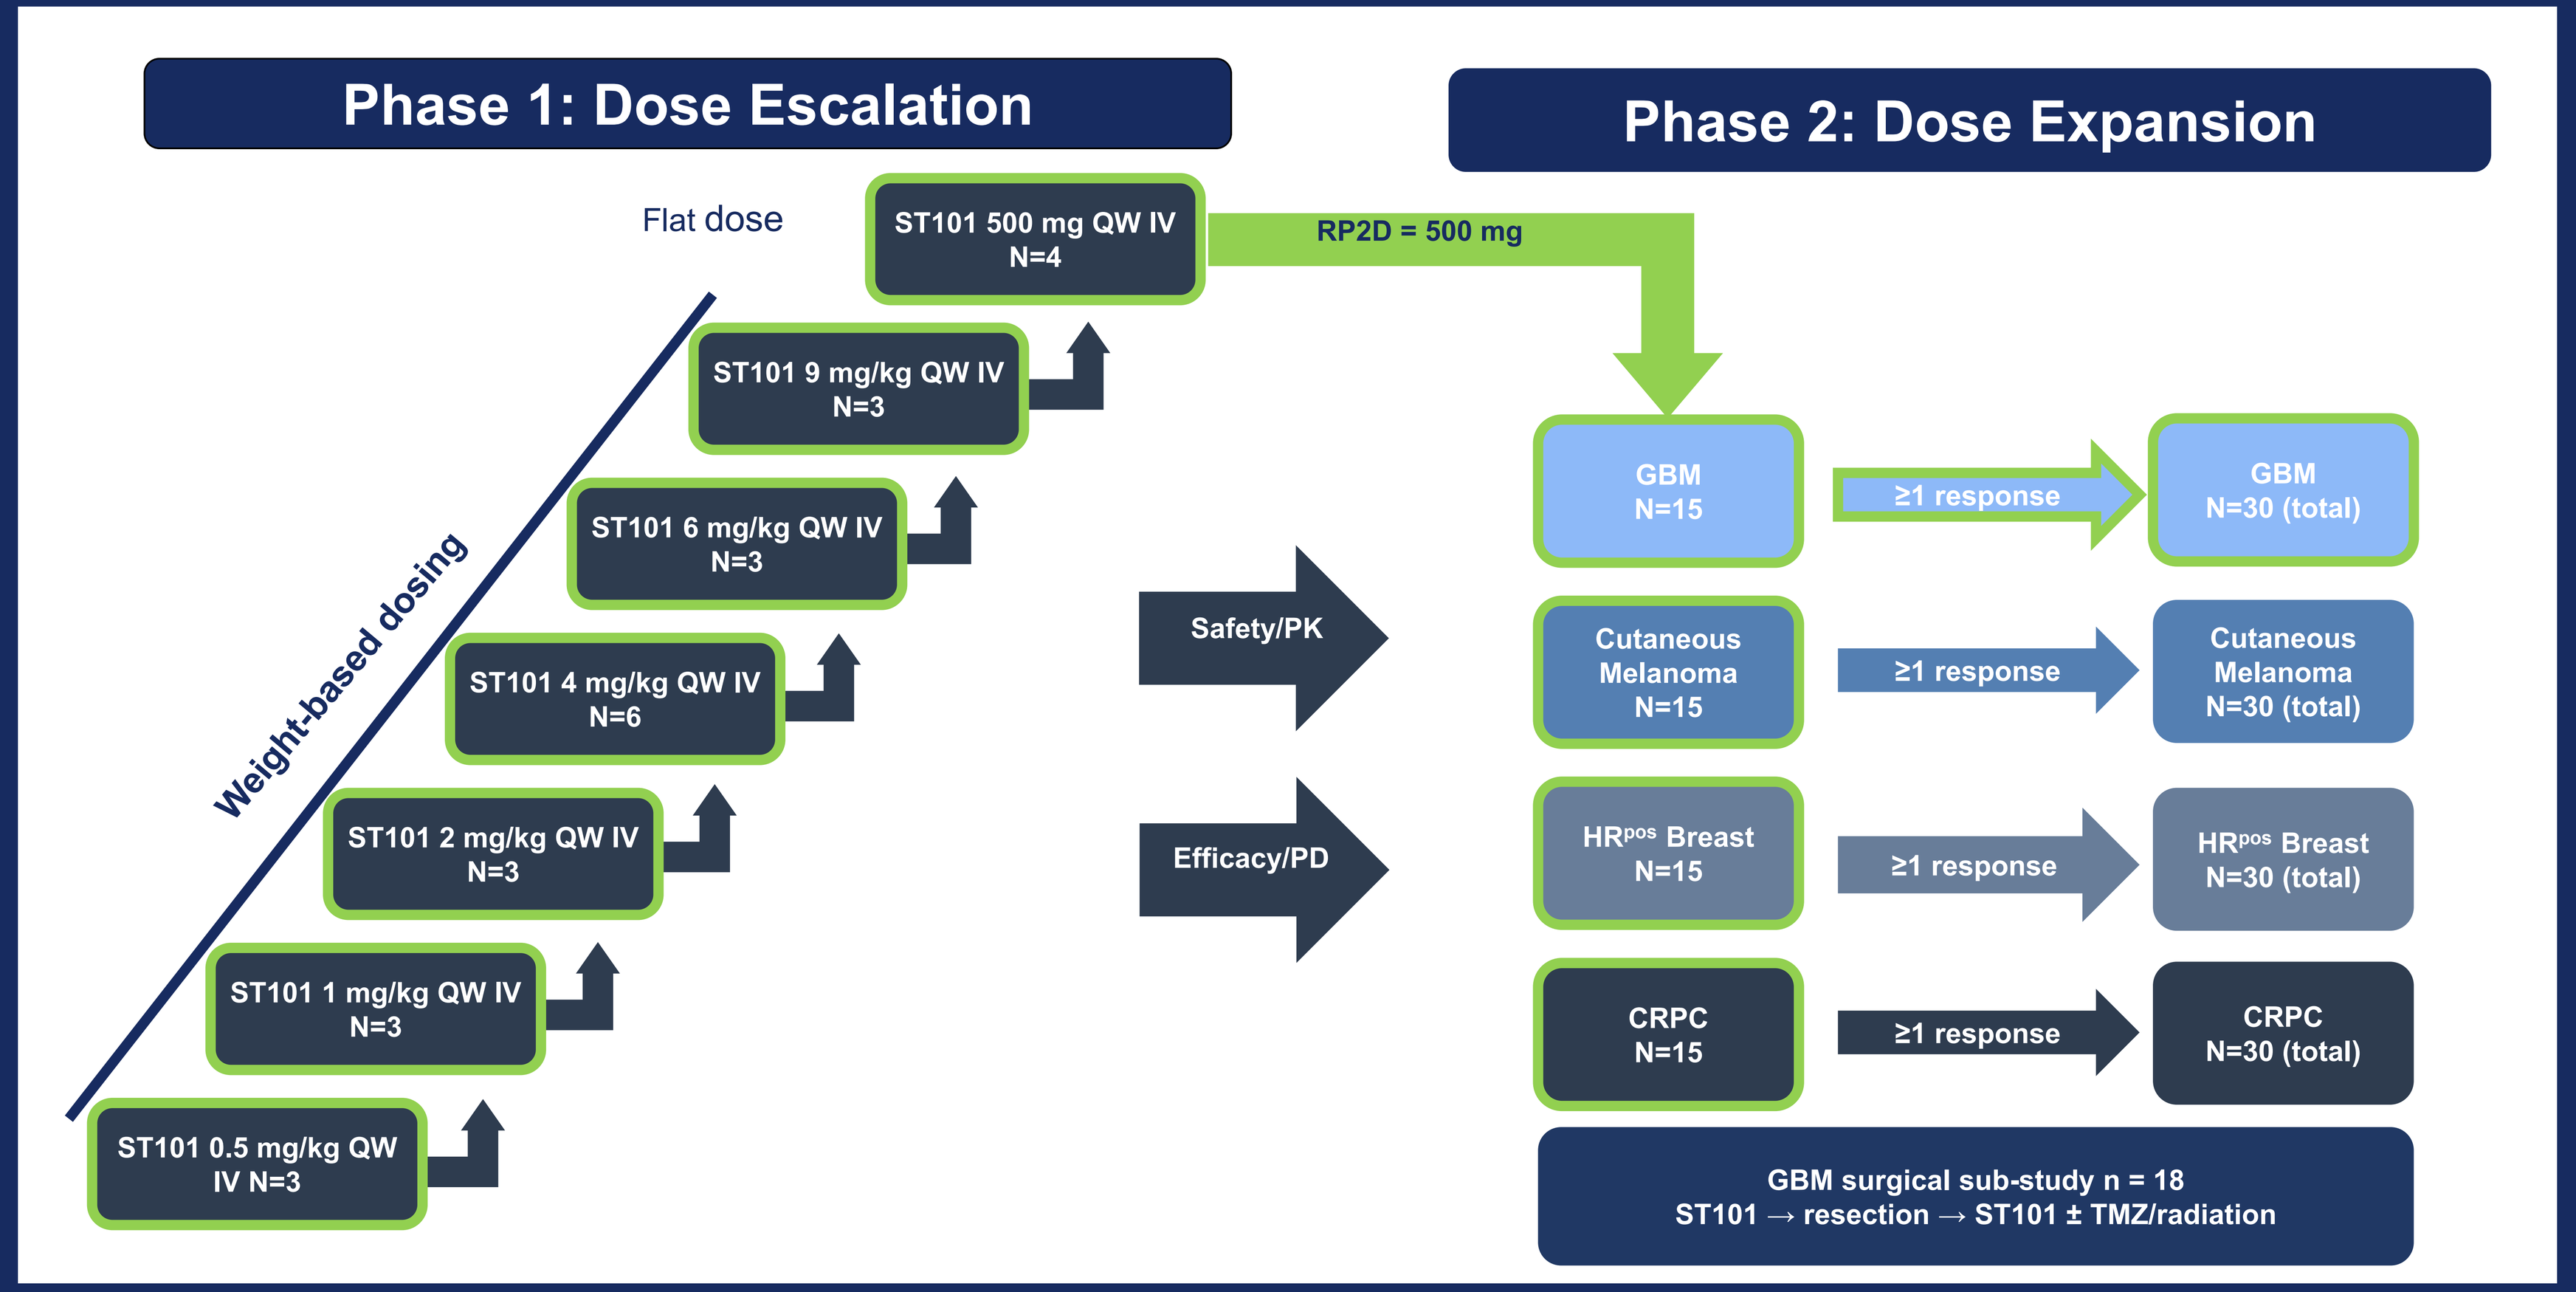
Phase 1: Dose Escalation
Phase 2: Dose Expansion
Flat dose
ST101 500 mg QW IV N=4
RP2D = 500 mg
ST101 9 mg/kg QW IV N=3
ST101 6 mg/kg QW IV N=3
ST101 4 mg/kg QW IV N=6
ST101 2 mg/kg QW IV N=3
ST101 1 mg/kg QW IV N=3
ST101 0.5 mg/kg QW IV N=3
Weight-based dosing
Safety/PK
Efficacy/PD
GBM N=15
≥1 response
GBM N=30 (total)
Cutaneous Melanoma N=15
≥1 response
Cutaneous Melanoma N=30 (total)
HRpos Breast N=15
≥1 response
HRpos Breast N=30 (total)
CRPC N=15
≥1 response
CRPC N=30 (total)
GBM surgical sub-study n = 18
ST101 → resection → ST101 ± TMZ/radiation

- 3+3 design involving 24 patients with advanced and metastatic solid tumors
- Primary objective - safety and tolerability of ST101
- Secondary objectives - preliminary efficacy in 4 tumor types, RP2D identification, PK, comparison of dosing regimens
- Exploratory objectives - investigate serum and tumor PD markers

Clinical benefit (SD,PR,CR)

Phase 1 (n=25)			
Tumor type	Dose	Response	Duration (weeks)
Signet ring adenoCa	0.5 mg/kg	SD	108
Small bowel adenoCa	2 mg/kg	SD	18
Abdominal sarcoma	2 mg/kg	SD	9
Cutaneous melanoma	4 mg/kg	PR	77+
Hepatocellular Ca	6 mg/kg	SD	18
Esophageal adenoCa	6 mg/kg	SD	9
Cutaneous melanoma	9 mg/kg	SD	53
Uveal melanoma	500 mg (flat dose)	SD	18
Mucosal melanoma	500 mg (flat dose)	SD	18

Phase 2 (n=44*)			
Tumor type	Dose	Response	Duration (weeks)
GBM	500 mg (flat dose)	PR	24+
GBM	500 mg (flat dose)	SD	13+
GBM	500 mg (flat dose)	SD	9+
Melanoma	500 mg (flat dose)	SD	18
Melanoma	500 mg (flat dose)	SD	9
Prostate	500 mg (flat dose)	SD	18
Prostate	500 mg (flat dose)	SD	9

*As of 10-Oct-22 17 patients pending first on-treatment scan

ST101 tumor uptake increases with dose

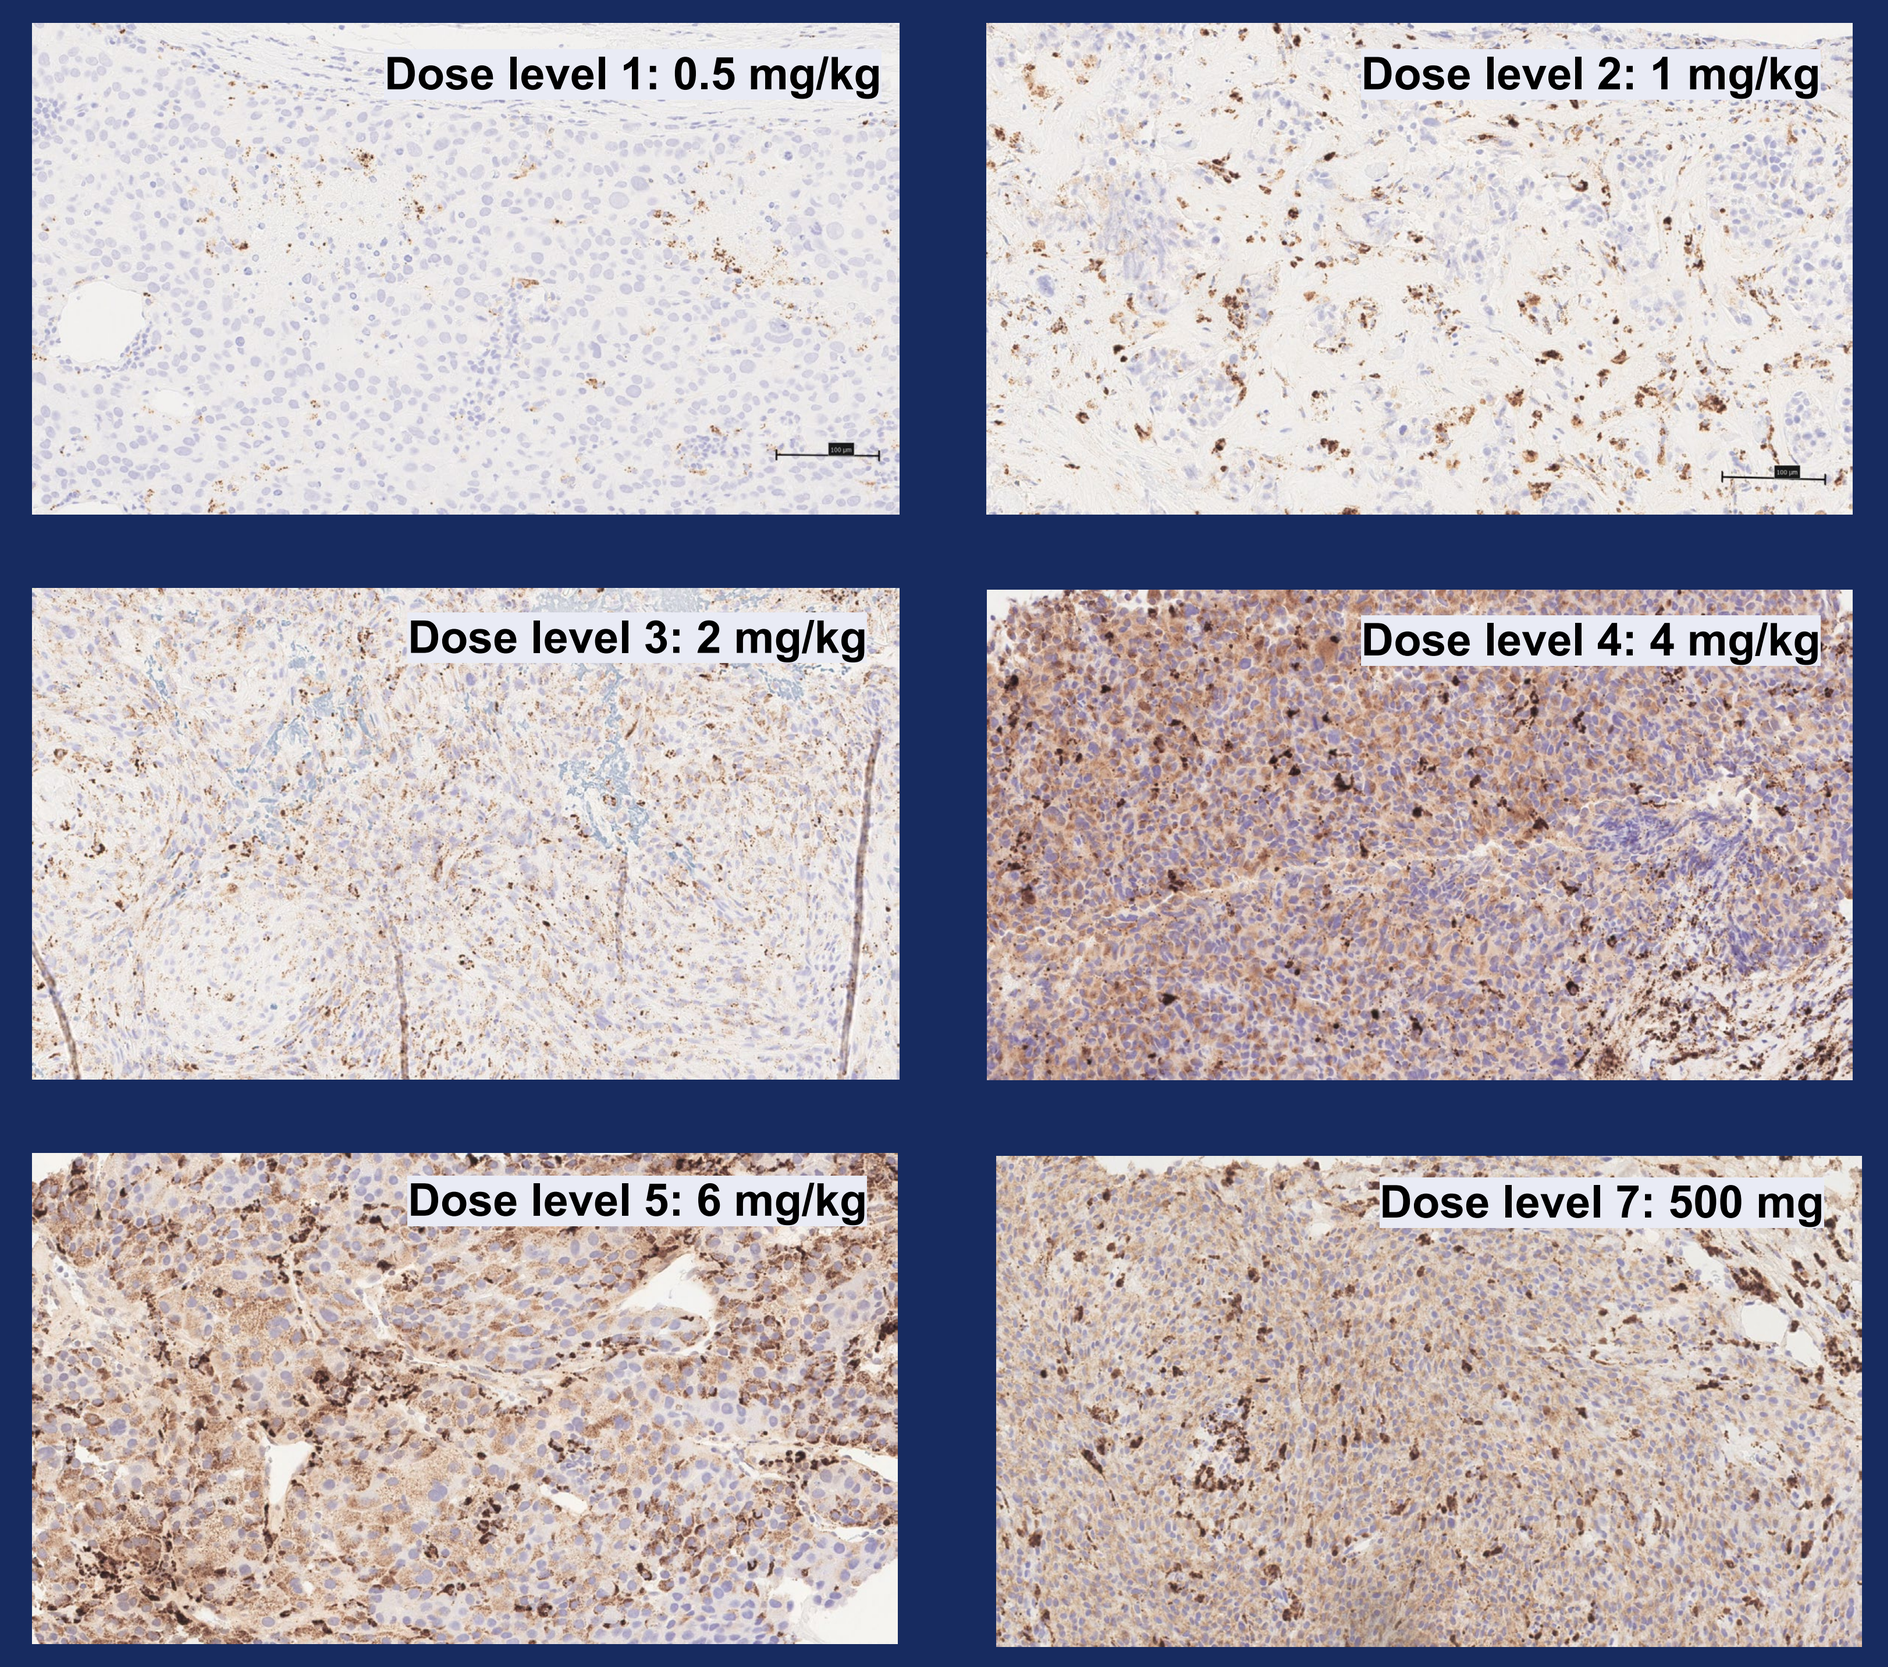
Dose level 1: 0.5 mg/kg
Dose level 2: 1 mg/kg
Dose level 3: 2 mg/kg
Dose level 4: 4 mg/kg
Dose level 5: 6 mg/kg
Dose level 7: 500 mg

ST101 decreases C/EBPβ expression

Baseline Cycle 2

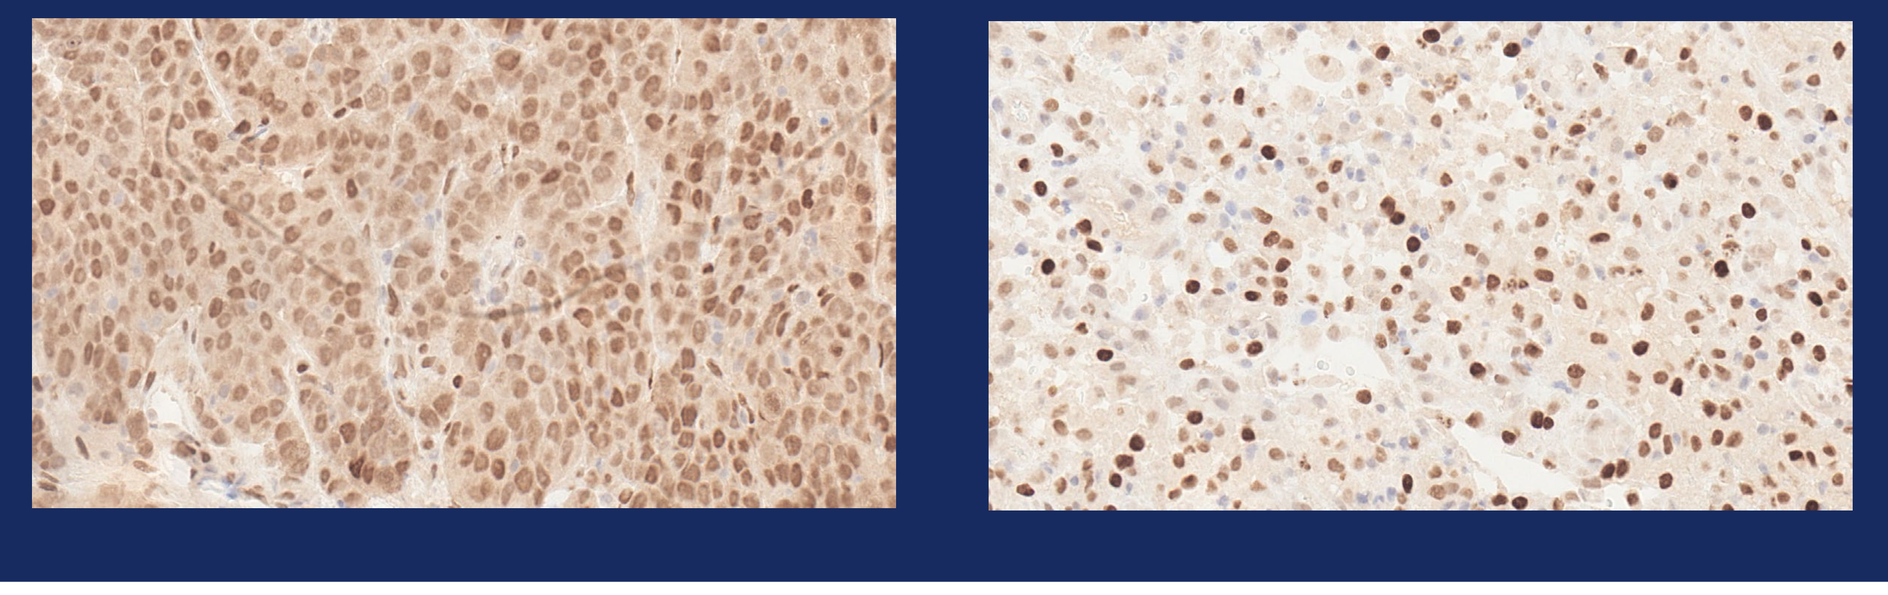

Cutaneous melanoma PR, phase 1 (4 mg/kg)

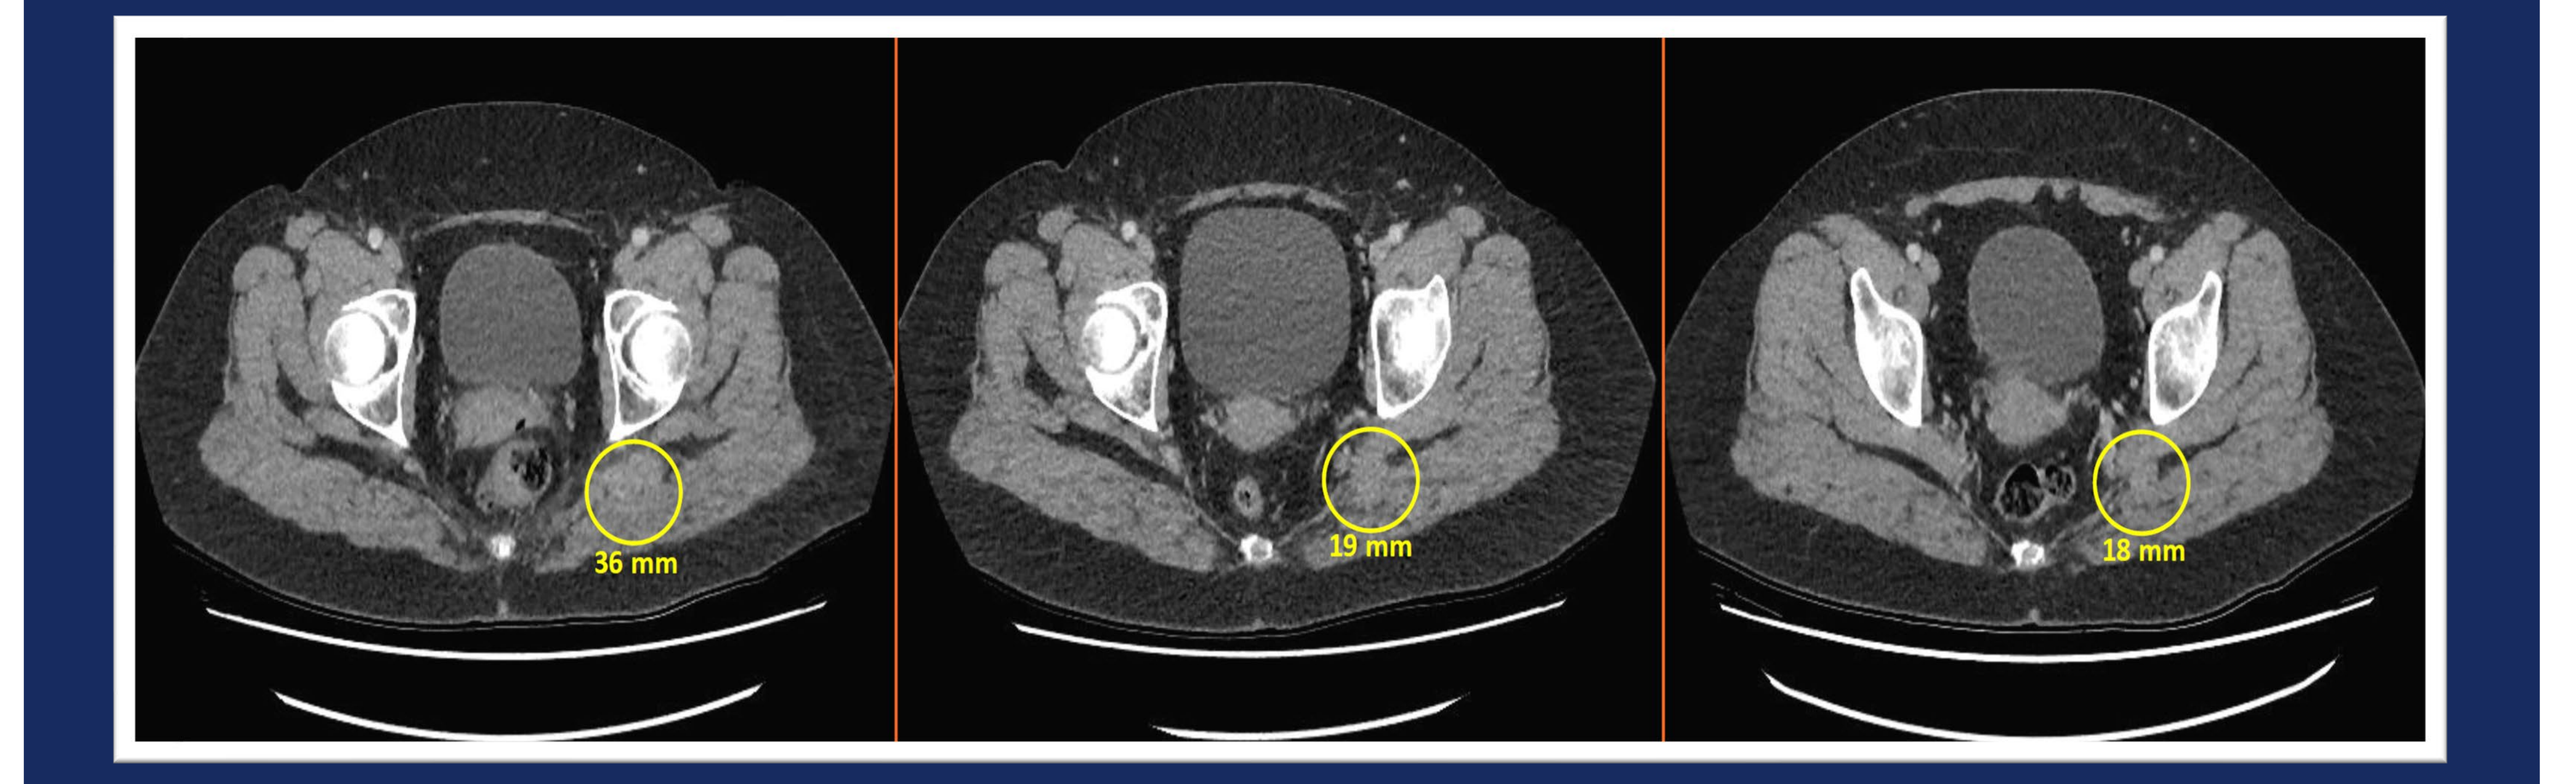
36 mm
19 mm
18 mm

Baseline Week 9 (Unconfirmed PR, -30%) Week 15 (Confirmed PR, -38%)

GBM PR, phase 2 (RP2D, 500 mg)

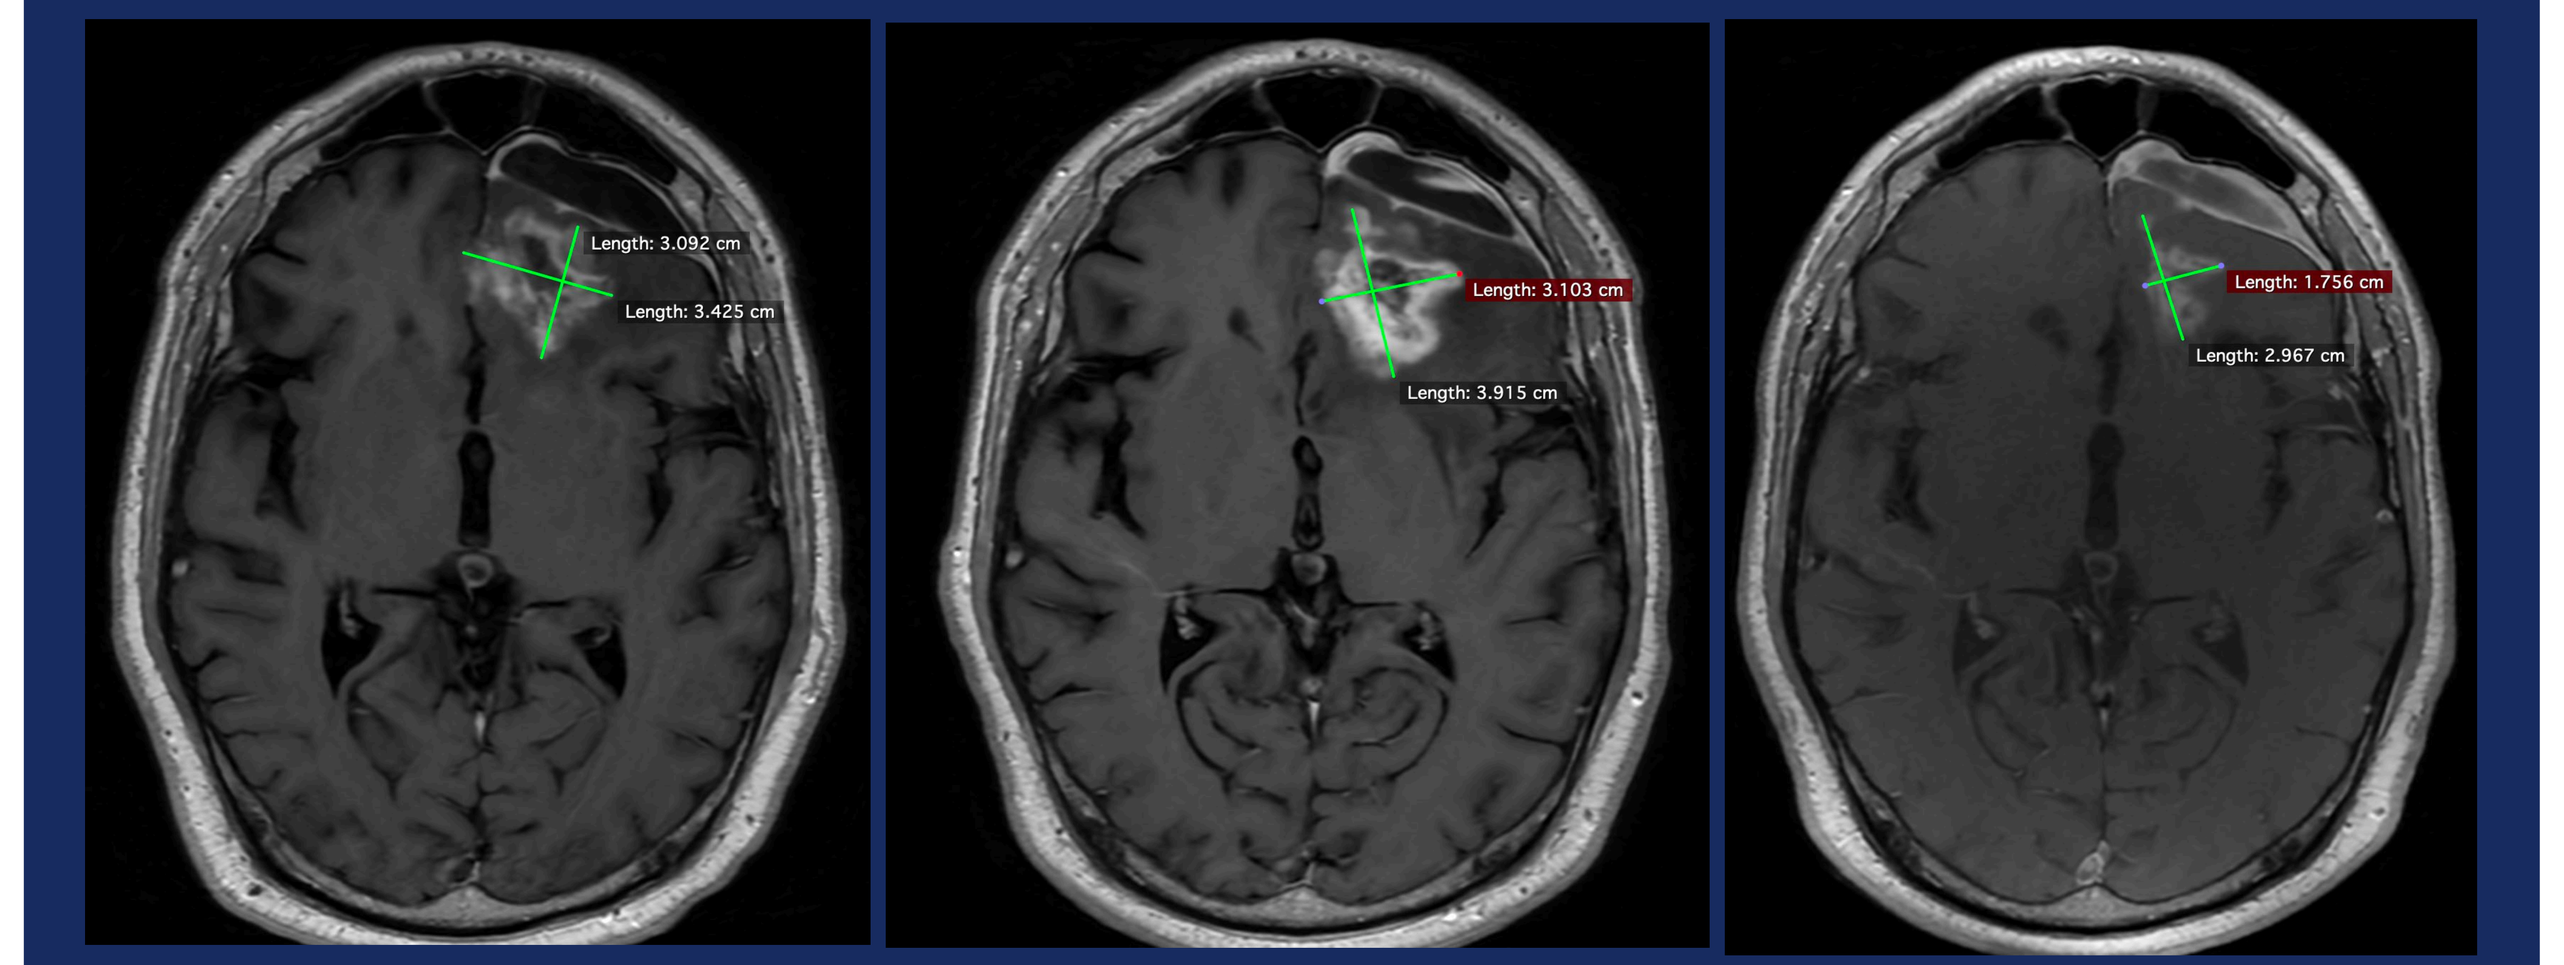
Length: 3.092 cm
Length: 3.425 cm
Length: 3.103 cm
Length: 3.915 cm
Length: 1.756 cm
Length: 2.967 cm

Baseline Week 4 (+14%) Week 14 (Unconfirmed PR, -50%)

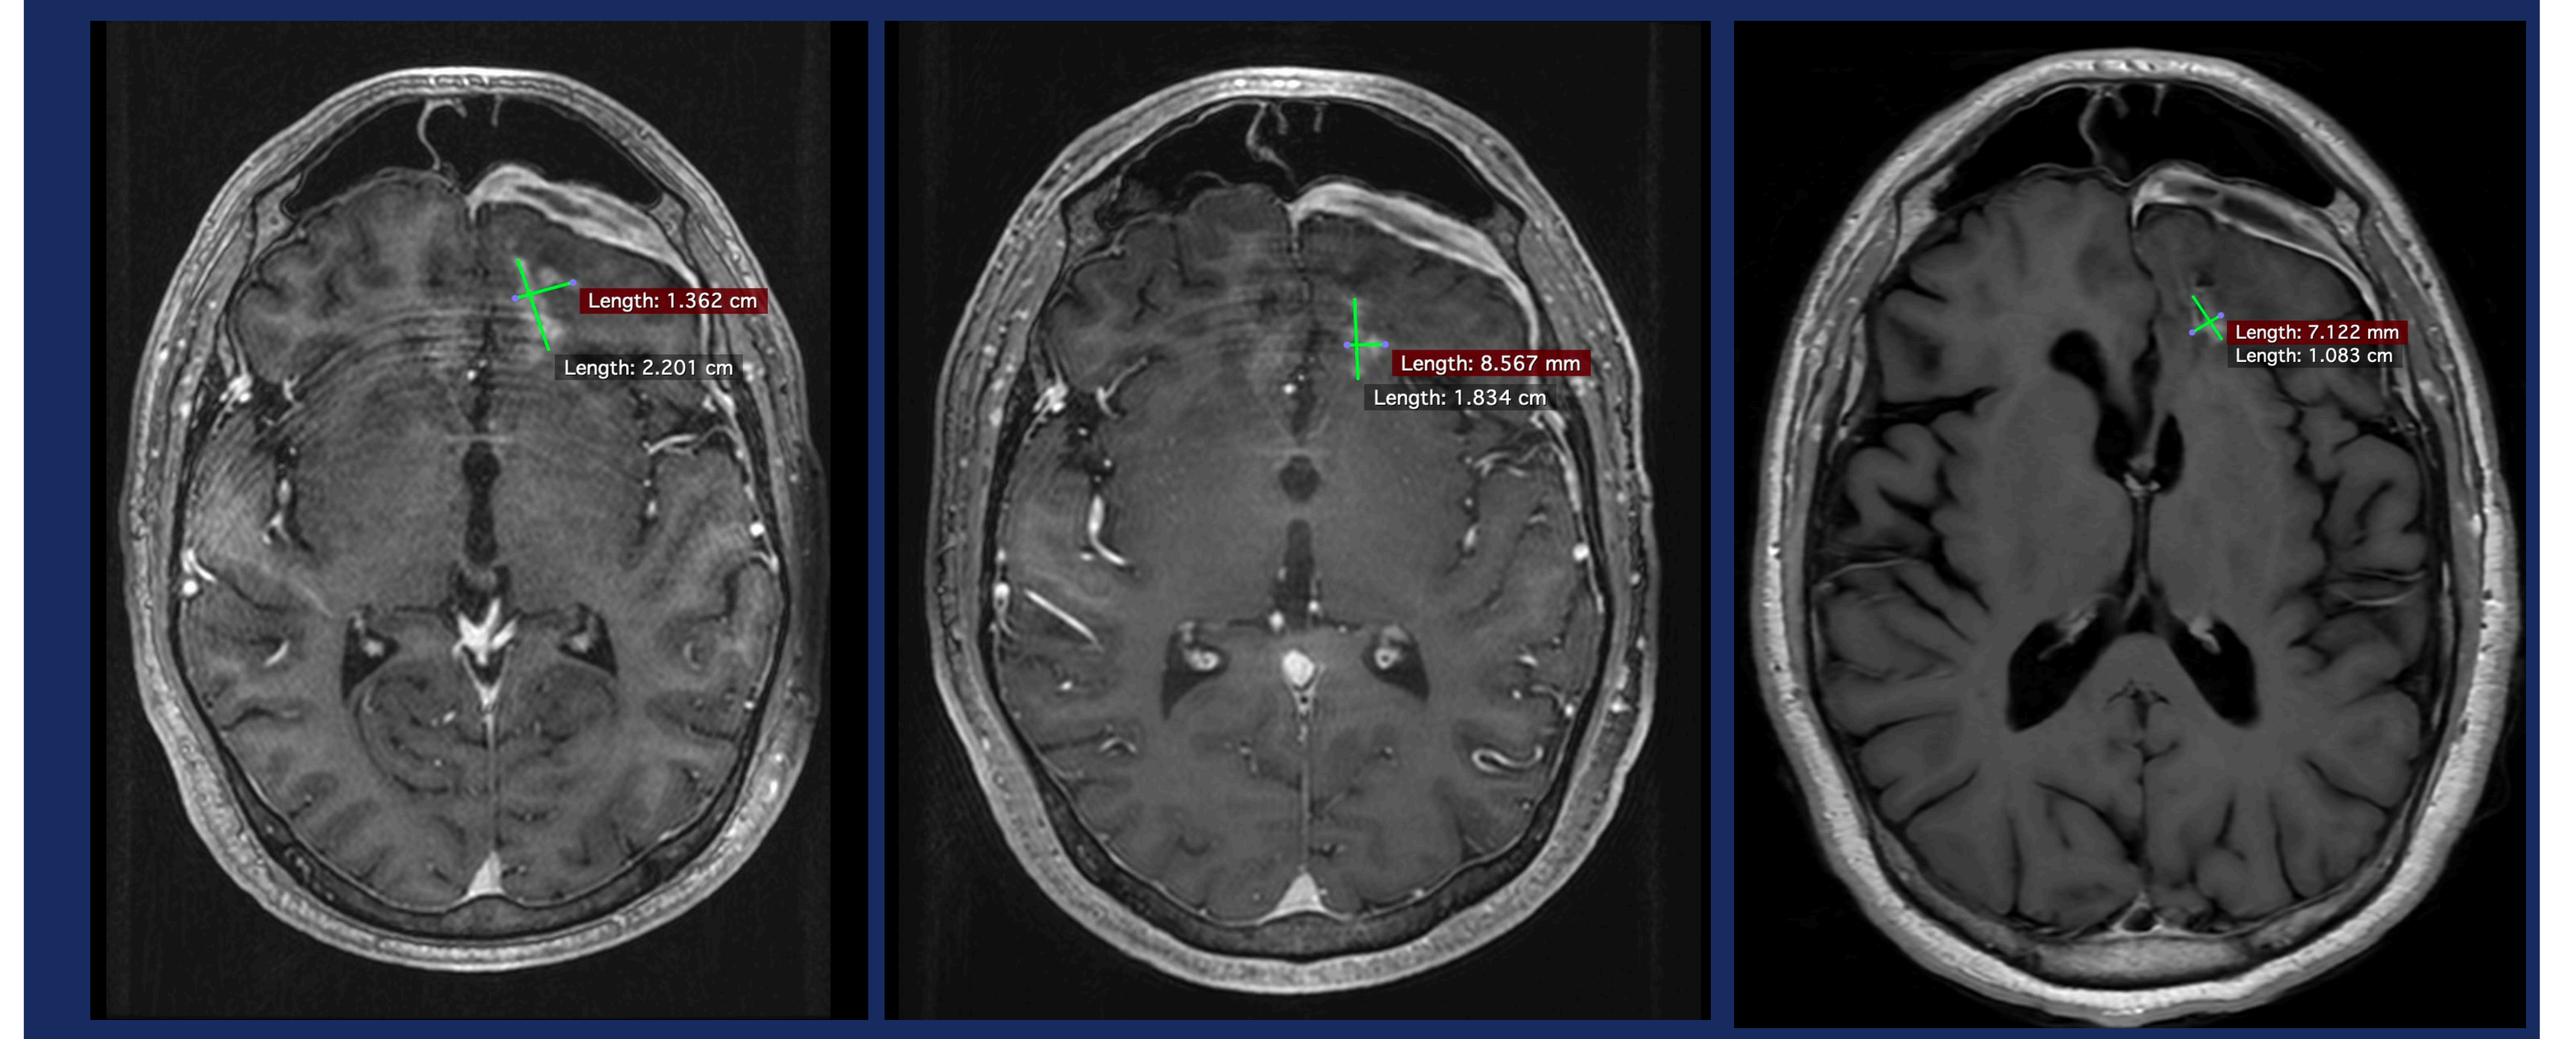
Length: 1.362 cm
Length: 2.201 cm
Length: 8.567 mm
Length: 1.834 cm
Length: 7.122 mm
Length: 1.083 cm

Week 18 (Confirmed PR, -71%) Week 27 (Sustained PR, -85%) Week 36 (Sustained PR, -93%)

Cutaneous melanoma SD, phase 1 (9 mg/kg)

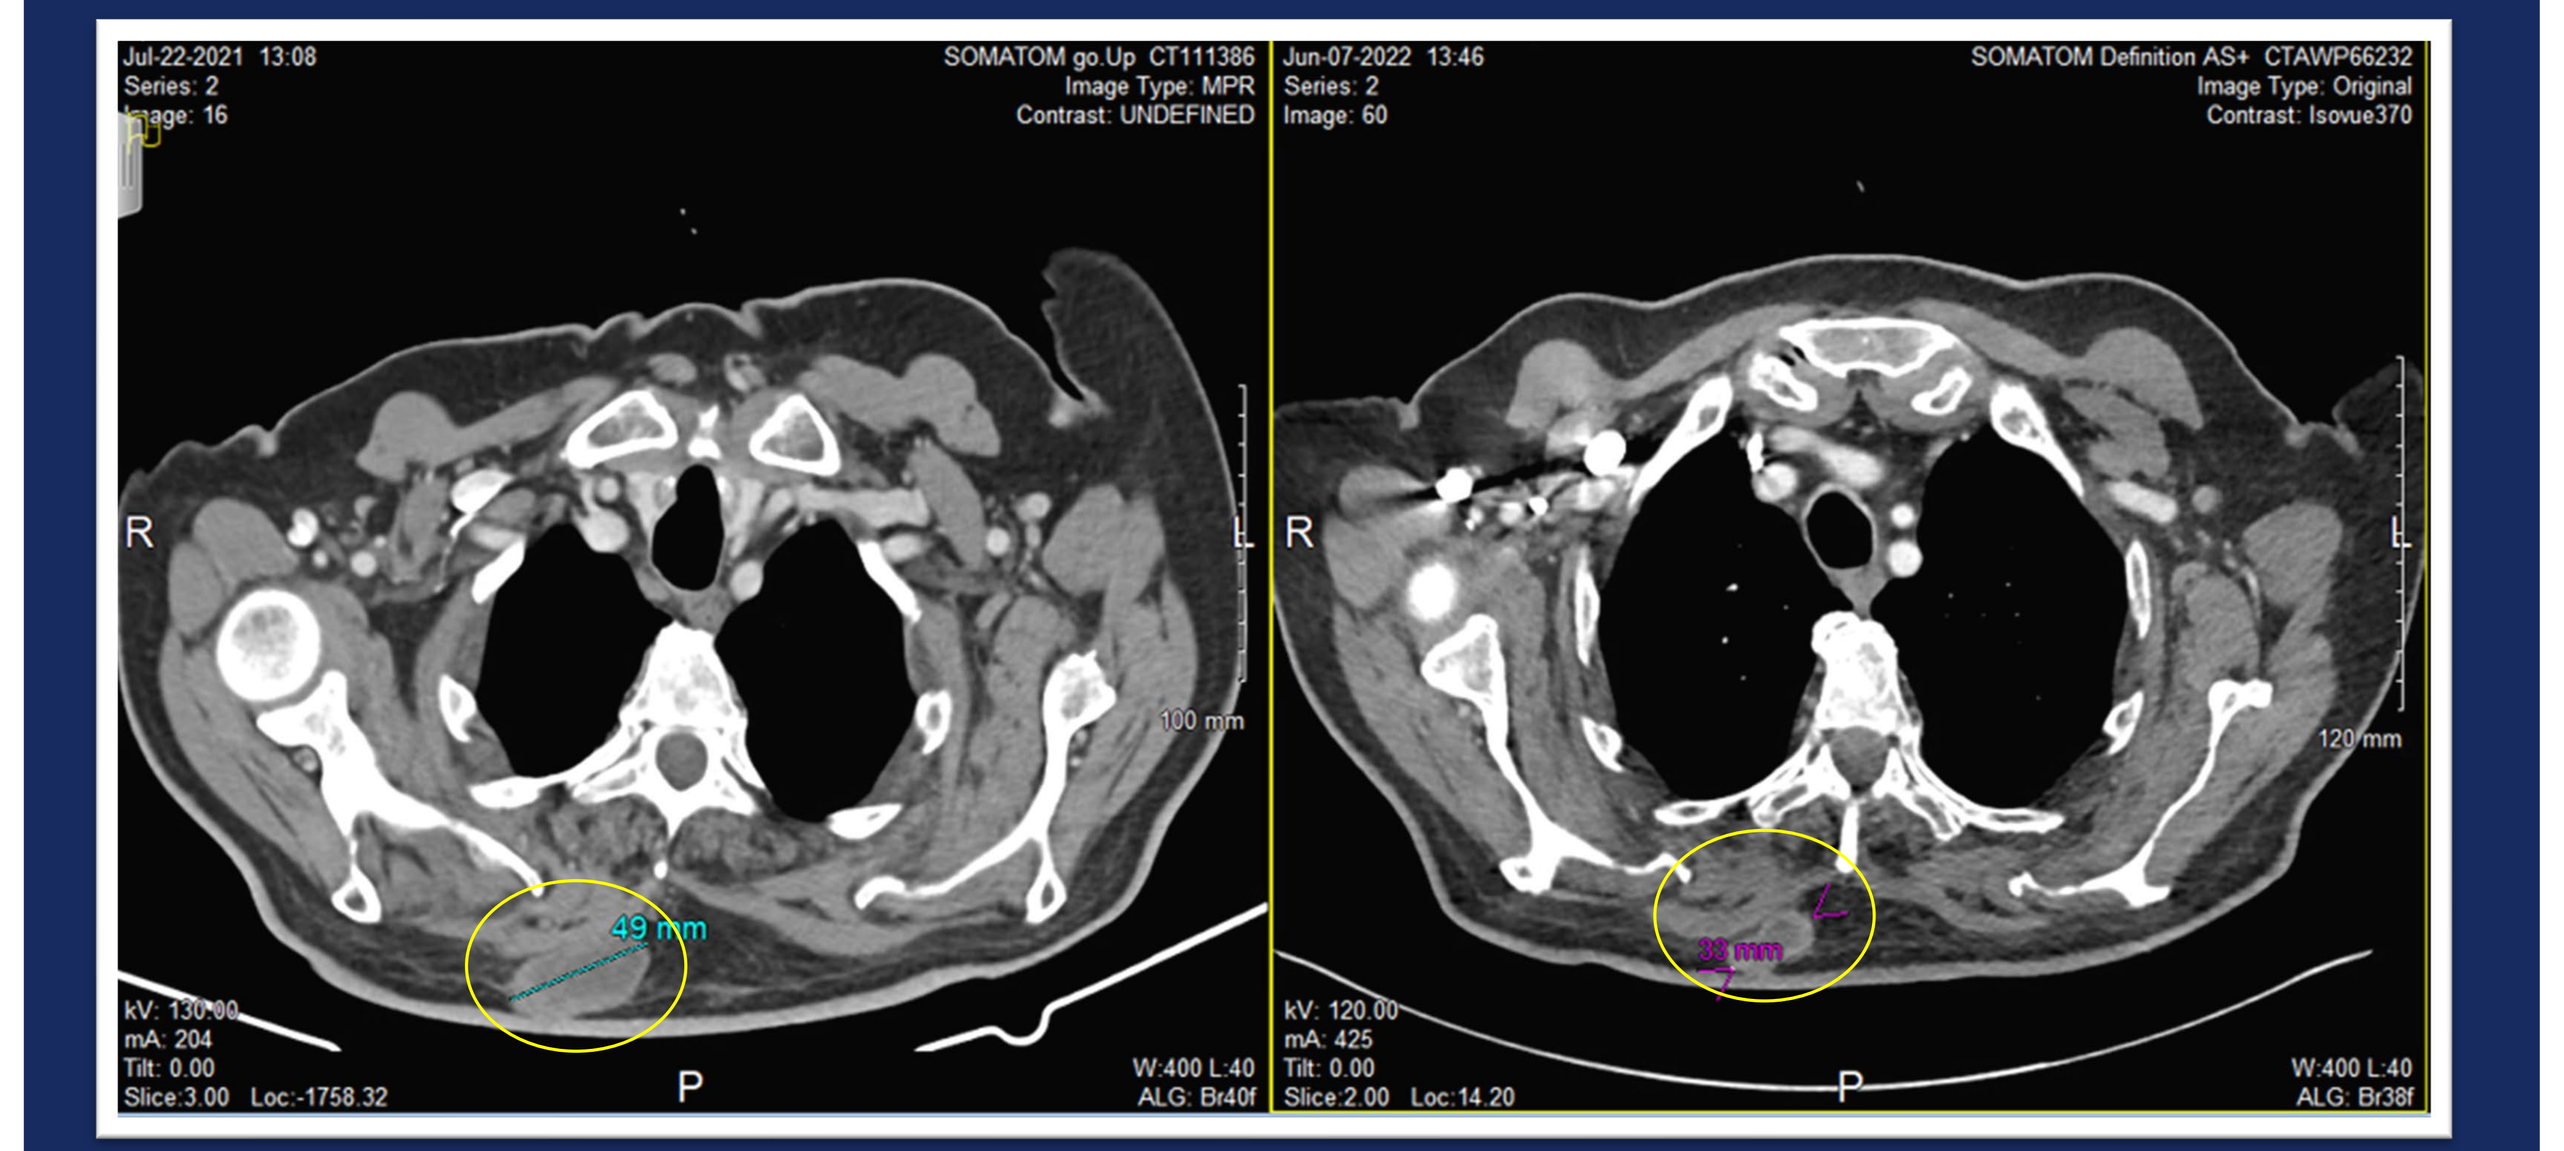
Jul-22-2021 13:08
Series: 2
Image: 16
SOMATOM go.Up CT111386
Image Type: MPR
Contrast: UNDEFINED
Jun-07-2022 13:46
Series: 2
Image: 60
SOMATOM Definition AS+ CTAWP66232
Image Type: Original
Contrast: Isovue370
R
L
P
49 mm
100 mm
120 mm
kV: 130.00
mA: 204
Tilt: 0.00
Slice:3.00 Loc:-1758.32
W:400 L:40
ALG: Br40f
kV: 120.00
mA: 425
Tilt: 0.00
Slice:2.00 Loc:14.20
W:400 L:40
ALG: Br38f

Baseline Week 44 (Stable disease, -23%)

No DLTs, no ST101-related SAEs and no withdrawal due to toxicity

TEAEs occurring in ≥ 10% of patients	n (69)	%
IRR symptoms	44	61
Blood creatinine increased	17	24
Fatigue	11	15
Nausea	10	14
Hypophosphatemia	7	10

IRRs decrease with subsequent infusions

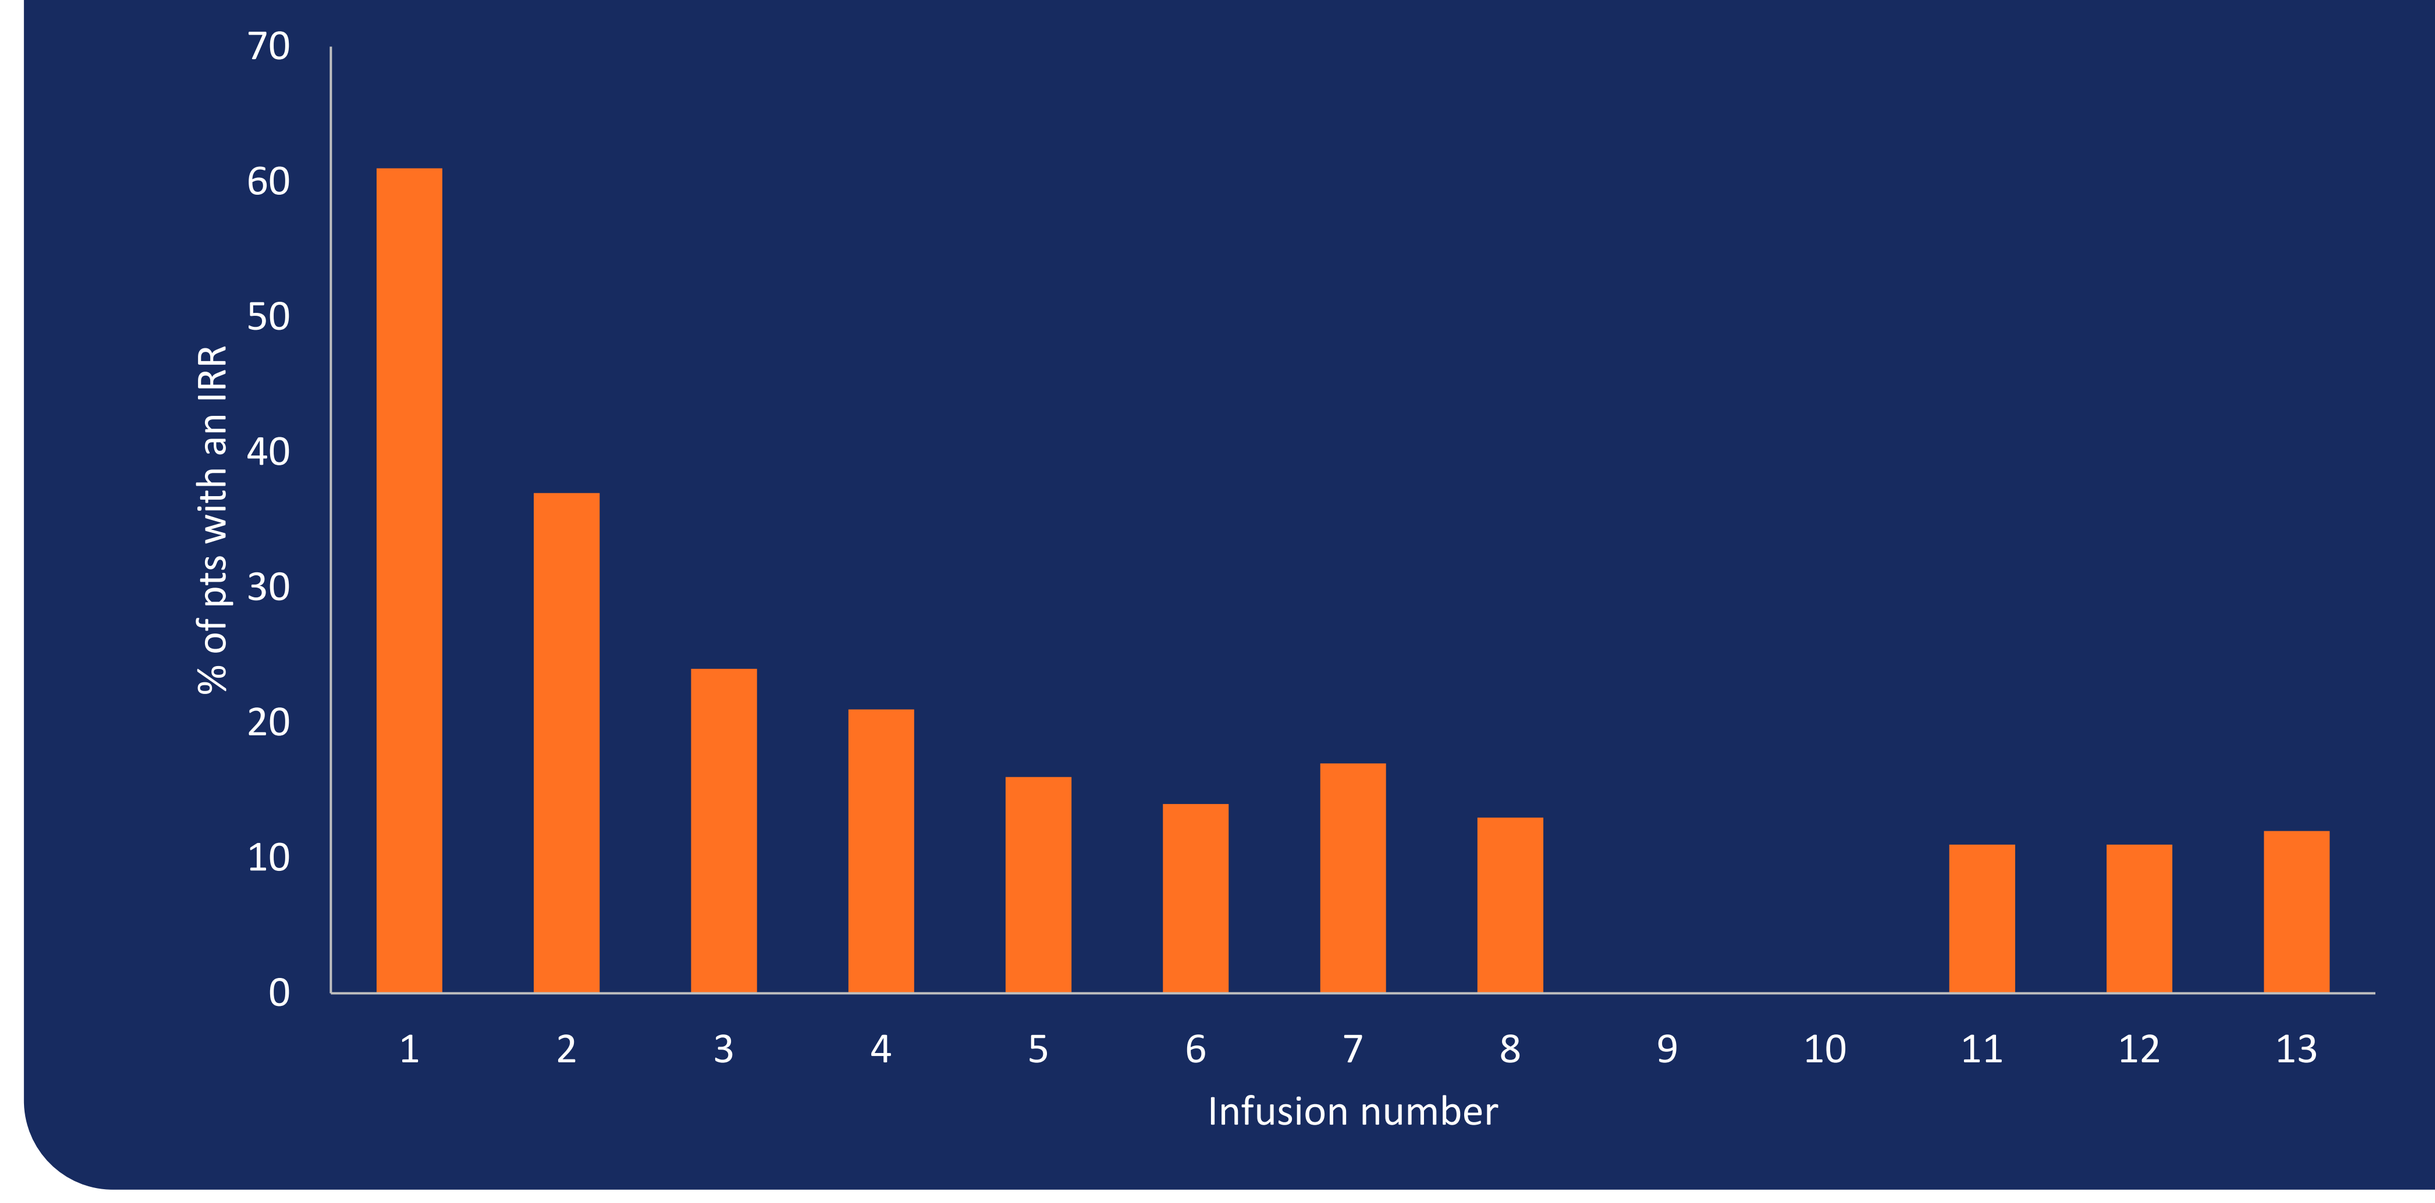
% of pts with an IRR
0
10
20
30
40
50
60
70
1
2
3
4
5
6
7
8
9
10
11
12
13
Infusion number

Conclusions

ST101 was safe and well tolerated

- Evidence of efficacy, particularly at higher doses
- No DLTs, related SAEs or withdrawals for toxicity
- IRRs were managed with simple standard approach
 - Interruption and slowing infusion rate
 - H1, H2 and leukotriene antagonists

Phase 1 clinical benefit

- 1 confirmed PR lasting > 77 weeks
- 8 patients with SD from 9 - 108 weeks

Phase 2 further efficacy signal

- 44 patients enrolled since the beginning of the year
- 1 GBM patient exhibiting a partial response for > 24 weeks
- 6 patients with stable disease (2 ongoing treatment)
 - 9 – 18 weeks in GBM, melanoma, and prostate cancers

Proof of concept achieved for this novel MOA and target

Sapiencetherapeutics.com
Confidential © 2022

Affiliations 1. Sarah Cannon Research Institute UK. 2. Sarah Cannon Research Institute at HealthONE, Denver, CO. 3. START Midwest, Grand Rapids 49546, MI. 4. University of Glasgow, Beatson West of Scotland Cancer Centre, Glasgow, UK. 5. Northwestern Medicine, Neuro-oncology, Chicago, USA. 6. Columbia University Irving Medical Center. 7. Sarah Cannon Research Institute, Tennessee Oncology, Nashville, TN. 8. Edinburgh Cancer Research Centre, University of Edinburgh, Edinburgh, UK. 9. University of California, Brain Tumor Center, San Francisco, USA. 10. Sapience Therapeutics Inc., Harrison, NY.

Abbreviations: AdenoCA = adenocarcinoma; ATF5 = activating transcription factor 5; BC = breast cancer; C/EBPβ = CCAAT/enhancer-binding protein β; CR = complete response; CRPC = castrate-resistant prostate cancer; DL = dose level; DLT = dose-limiting toxicity; GBM = glioblastoma multiforme; HRpos = hormone receptor positive; IRR = infusion-related reaction; IV = intravenous; MOA = mechanism of action; PD = pharmacodynamic; PK = pharmacokinetic; PR = partial response; QW = once weekly dosing; RP2D = recommended phase 2 dose; SAE = serious adverse event; SD = stable disease; TEAEs = treatment-emergent adverse events, TMZ=temozolomide

References: 1. van de Vijveret et al., NEJM 2002; 2. Woboda et al., Oncogene 2020; 3. Healy et al., Epigenetics 2010; 4. Adamo et al., Prostate 2018; 5. Homma et al., Oncology Reports (2006), Vol 15; 6. Marigo (2010) Immunity; 7. Lampkin (2019) Brain, Behavior and Immunity