Low Incidence of Events with Oliceridine in Patients Predicted at High Risk for Developing Opioid-Induced Respiratory Depression

3001

Ashish K. Khanna[1], Cathy Michalsky[2], Kanaka Sridharan[2], Erik DeBoer[2], Sabry Ayad[3]
[1]Wake Forest School of Medicine, Winston-Salem, NC, USA; [2]Trevena, Inc. Chesterbrook, PA, USA; [3]Cleveland Clinic, Outcomes Research, Anesthesiology Institute, Fairview Hospital, Cleveland, OH, USA

ASRA PAIN MEDICINE
47th Annual Regional Anesthesiology & Acute Pain Medicine Meeting
March 31-April 2, 2022 | Las Vegas, NV | #ASRASPRING22

INTRODUCTION

- In the management of acute postoperative pain, IV opioids remain an important pharmacotherapy; however, the benefits of analgesic effects are limited by opioid-related adverse events (ORAEs).[1]
- Opioid-induced respiratory depression (OIRD) is among the most serious of the ORAEs that increase perioperative cost and hospital length of stay (LOS) and may result in permanent morbidity and mortality.[2,3]
- The prospective, observational trial of blinded continuous capnography and oximetry, PRediction of Opioid-induced respiratory Depression In patients monitored by capnoGraphY (PRODIGY) trial that investigated the incidence and risk factors associated with OIRD episodes in hospitalized patients receiving parenteral opioids, reported a 46% incidence of respiratory depression.[4]
- A respiratory depression episode in PRODIGY was defined as respiratory rate ≤ 5 breaths/min (bpm), oxygen saturation ≤ 85%, or end-tidal carbon dioxide ≤ 15 or ≥ 60 mm Hg for ≥ 3 minutes; apnea episode lasting > 30 seconds; or any respiratory opioid-related adverse event.
- The PRODIGY trial aided in creating a validated novel respiratory depression risk prediction tool, including 5 easy-to-assess variables: age ≥ 60 years by decade, sex, opioid naïvety, sleep disorders, and chronic heart failure.
- Oliceridine is a biased G-protein selective, IV opioid analgesic at μ-opioid receptors,[5] indicated in adults for the management of acute pain severe enough to require an intravenous opioid analgesic and for whom alternative treatments are inadequate.
- The G-protein selectivity results in analgesia with less recruitment of β-arrestin, a signaling pathway associated with adverse events such as OIRD.[5]
- Preclinical studies report that oliceridine is potently analgesic while causing less gastrointestinal dysfunction and respiratory suppression than morphine at equianalgesic doses.[5] The translation of these nonclinical findings into clinical results in humans has not been fully established.
- Findings from an exploratory analysis of a Phase 3 open-label, multicenter safety study, ATHENA, reported that the postoperative use of IV oliceridine in patients with advanced age and/or increased body mass index (≥ 30 kg/m^2) for the treatment of moderate-to-severe pain, was not associated with increased risk of OIRD. OIRD was defined by a) use of naloxone; b) respiratory rate < 10 bpm; or c) oxygen saturation (SpO_2 < 90%) within 48 hours of last dose of oliceridine.[6]
- The ATHENA, open-label safety study, did not use continuous capnography and oximetry to monitor patients for OIRD.
- In this exploratory analysis from the ATHENA trial (**Figure 1**), we applied the PRODIGY risk-scoring tool in patients with pre-existing comorbidities of sleep apnea and/or chronic heart failure to categorize them into low-, intermediate-, or high-risk and report the incidence of OIRD in these patients.

METHODS

- PRODIGY scoring utilized in the exploratory analysis is shown in **Table 1**.
- The sum of points categorized patients to low (< 8 points), intermediate (≥ 8 and < 15 points), and high (≥ 15 points) risk for development of OIRD.
- The incidence of OIRD in these 3 predicted risk categories of patients using the definition of OIRD as reported in the previous analysis from ATHENA is reported.

DISCLOSURES AND ACKNOWLEDGEMENTS

- ATHENA Study: NCT02656875.
- Layout for the poster was provided by Innovation Communications Group.

Figure 1: ATHENA Study Design and Treatment Protocol

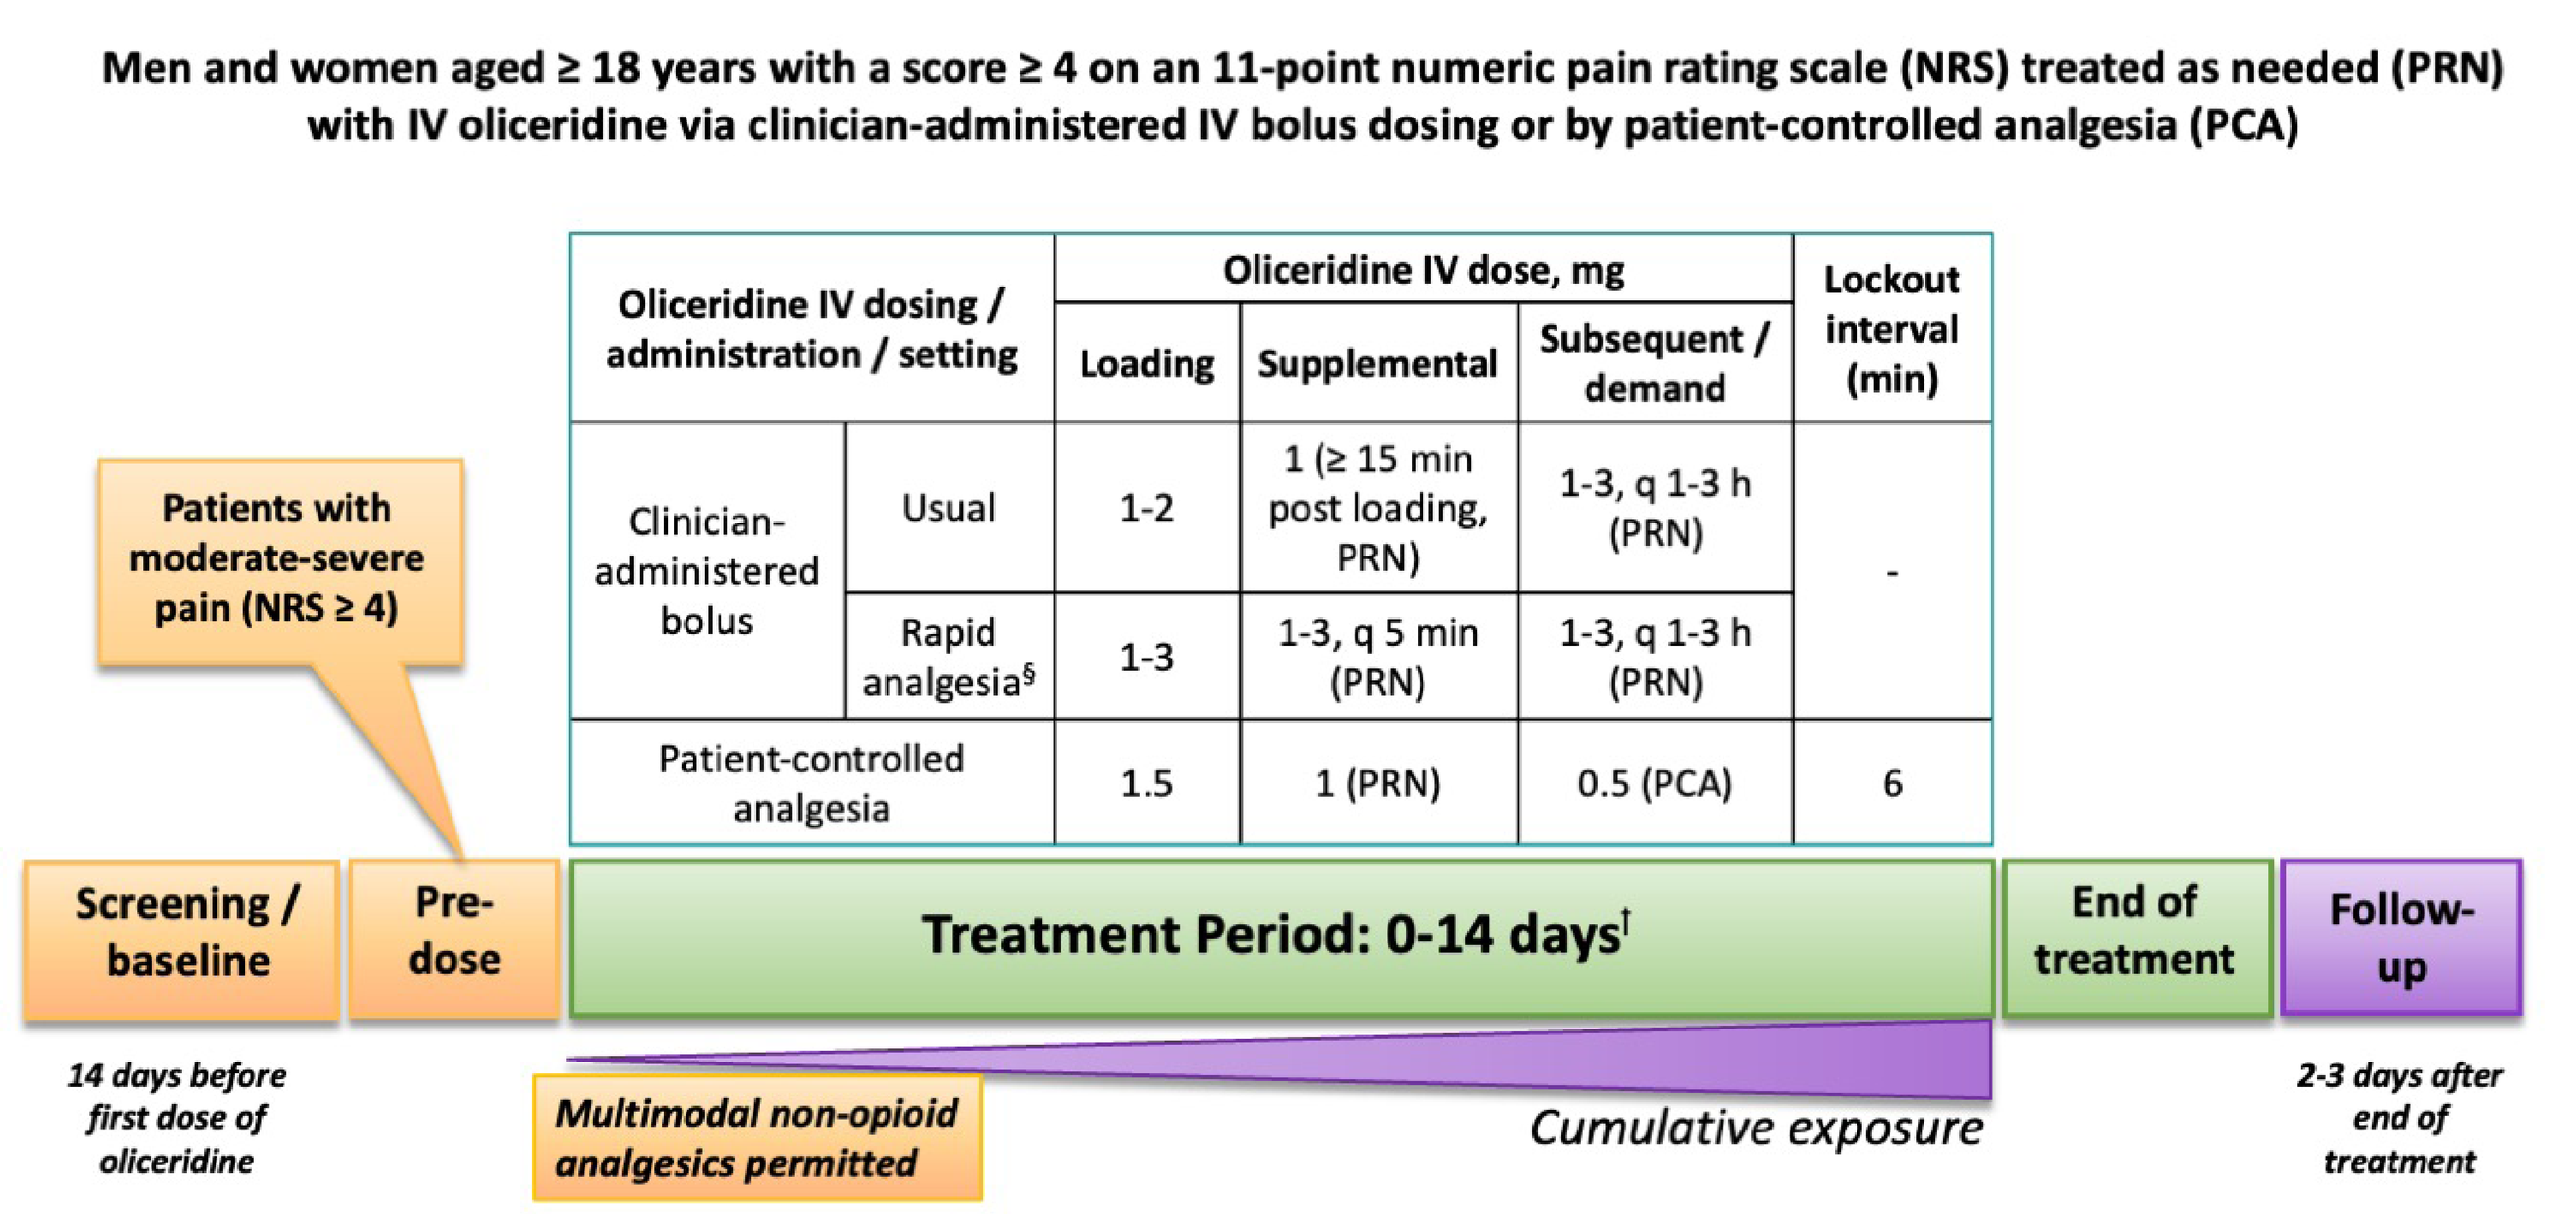
Men and women aged ≥ 18 years with a score ≥ 4 on an 11-point numeric pain rating scale (NRS) treated as needed (PRN) with IV oliceridine via clinician-administered IV bolus dosing or by patient-controlled analgesia (PCA)
Oliceridine IV dosing / administration / setting
Oliceridine IV dose, mg
Loading
Supplemental
Subsequent / demand
Lockout interval (min)
Clinician-administered bolus
Usual
1-2
1 (≥ 15 min post loading, PRN)
1-3, q 1-3 h (PRN)
-
Rapid analgesia§
1-3
1-3, q 5 min (PRN)
1-3, q 1-3 h (PRN)
Patient-controlled analgesia
1.5
1 (PRN)
0.5 (PCA)
6
Patients with moderate-severe pain (NRS ≥ 4)
Screening / baseline
Pre-dose
Treatment Period: 0-14 days†
End of treatment
Follow-up
14 days before first dose of oliceridine
Multimodal non-opioid analgesics permitted
Cumulative exposure
2-3 days after end of treatment

§Settings in which rapid analgesia were required, e.g., emergency department or post-anesthesia care unit (PACU)
†As determined by the clinical need for parenteral opioid therapy
IV=intravenous; NRS=numeric rating scale; PRN, as needed

Table 1: PRODIGY Risk Score Utilization in ATHENA Patients with Comorbidities of Sleep Apnea and/or Chronic Heart Failure*

Clinical Characteristic	Points if clinical characteristic is "Yes" (Khanna et al. 2020)[4]
Age, years	
< 60	0
≥ 60 - < 70	8
≥ 70 - < 80	12
≥ 80	16
Sex (M)	8
Opioid naïve*	3
Sleep apnea*	5
Chronic heart failure (including congestive heart failure)*	**7**

*For this exploratory analysis of the ATHENA trial, we utilized the PRODIGY risk-scoring tool using the variables age ≥ 60 years by decade, sex, and opioid naïvety based on whether the patient had any prior medication within the opioid class (excluding opioid used perioperatively) in the WHO dictionary coding and applied in patients who had a medical history of sleep apnea and/or chronic heart failure (including congestive heart failure). Prior medication in the ATHENA study was defined as medications administered within the 14 days before the first dose of oliceridine.

Table 2: PRODIGY Score Distribution in the ATHENA Patients with Comorbidities of Sleep Apnea and/or Chronic Heart Failure

	Low Risk	Intermediate Risk	High Risk
PRODIGY SCORE	< 8 points	≥ 8 to < 15 points	≥ 15 points
N	**16/103 (15.5%)**	**47/103 (45.6%)**	**40/103 (38.8%)**

Table 3: Clinical Characteristics of Patients in The Three Predicted Risk Categories

	Low Risk	Intermediate Risk	High Risk
PRODIGY SCORE	< 8 points	≥ 8 to < 15 points	≥ 15 points
N	**16**	**47**	**40**
Age, mean (SD) years, n (%)			
< 60, years	16 (100.0)	25 (53.2)	1 (2.5)
60 — < 70, years	-	22 (46.8)	19 (47.5)
70 — < 80, years	-	-	17 (42.5)
≥ 80, years	-	-	3 (7.5)
Sex			
Male, n (%)	-	24 (51.1)	31 (77.5)
Female, n (%)	16 (100.0)	23 (48.9)	9 (22.5)
BMI, mean (SD) kg/m^2	**43.9 (10.5)**	**34.5 (6.6)**	**33.0 (5.5)**
Opioid naïve, n (%)	0	0	0
Sleep apnea, n (%)	16 (100.0)	47 (100.0)	34 (85.0)
Chronic heart failure , n(%)	0	1 (2.1)	9 (22.5)
Baseline pain score, n (%)	7.1 (3.2)	6.4 (2.3)	5.4 (1.7)

BMI=Body mass index

Table 4: Oliceridine Dose and Duration of Treatment

	Low Risk	Intermediate Risk	High Risk
PRODIGY SCORE	< 8 points	≥ 8 to < 15 points	≥ 15 points
N	**16**	**47**	**40**
Oliceridine Dose, mg			
Mean (SD)	34.6 (28.0)	31.9 (28.8)	31.4 (32.7)
Range	(1.0, 104.0)	(1.0, 114.0)	(1.0, 158.9)
Duration of Treatment, hours			
Mean (SD)	24.7 (20.4)	28.2 (24.6)	33.8 (29.9)
Range	(0, 69.7)	(0, 76.8)	(0, 130.8)

Figure 2: Incidence of OIRD in the Three Predicted Risk Categories

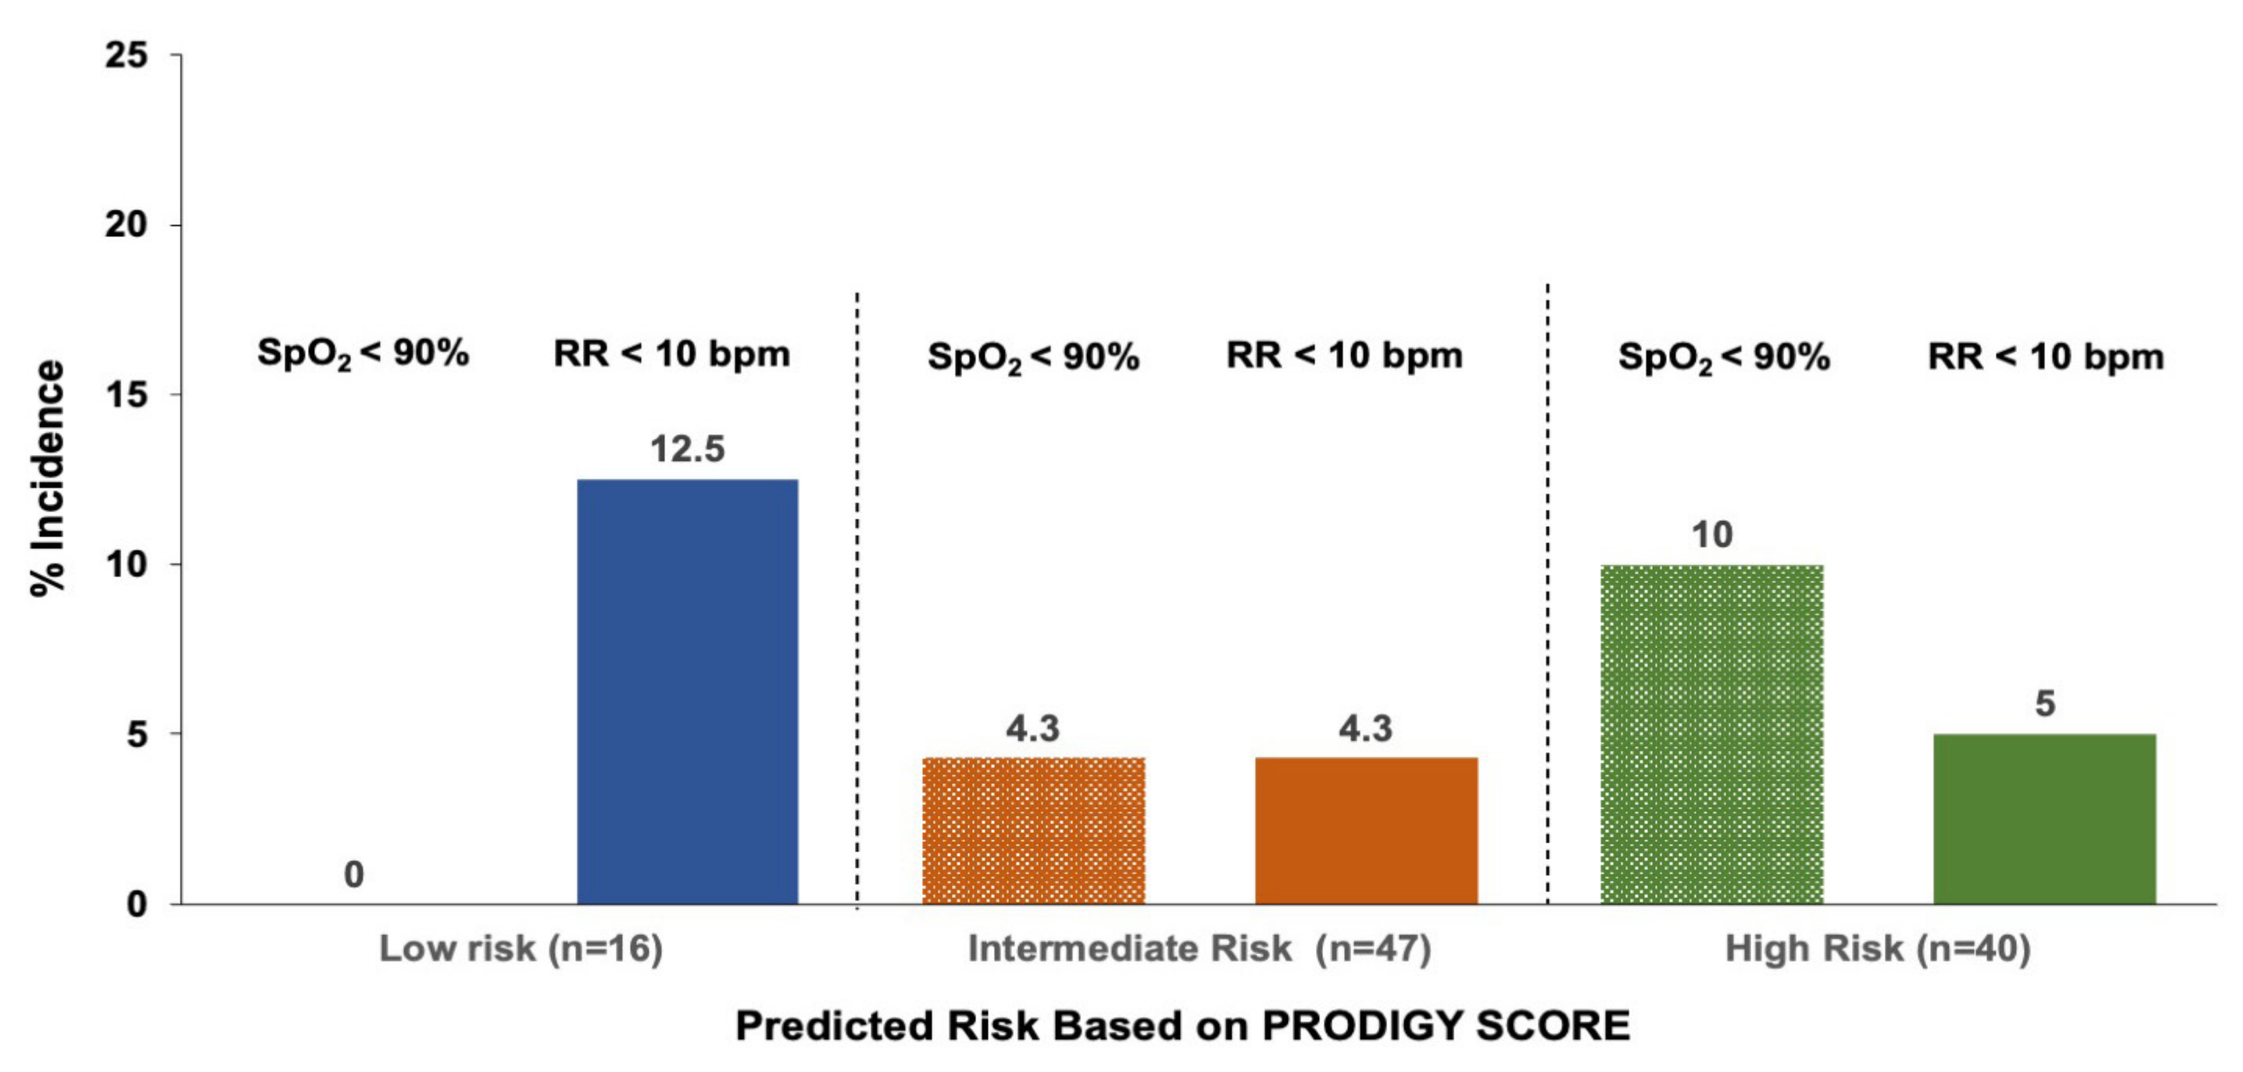
25
20
15
10
5
0
% Incidence
SpO2 < 90%
RR < 10 bpm
SpO2 < 90%
RR < 10 bpm
SpO2 < 90%
RR < 10 bpm
0
12.5
4.3
4.3
10
5
Low risk (n=16)
Intermediate Risk (n=47)
High Risk (n=40)
Predicted Risk Based on PRODIGY SCORE

SpO_2 <90% = oxygen saturation <90%; RR = respiratory rate; bpm = beats per minute.

RESULTS

- A total of 768 patients (mean age 54.1 ± 16.1 y; 65% females) were treated with oliceridine (up to 6 days).
- 103 patients had a medical history of sleep apnea and/or chronic heart failure.
 - Applying the PRODIGY risk-scoring tool in patients with the medical comorbidities of sleep apnea and/or chronic heart failure, most patients were predicted to be of "intermediate-risk" or "high-risk" for the development of OIRD (**Table 2**).
- Clinical characteristics of patients in the three predicted risk categories are shown in **Table 3**. None of the patients were opioid naïve.
- The dose of oliceridine, and duration of exposure is shown in **Table 4**.
 - Patients in the intermediate- or high-risk categories had a longer duration of treatment than patients in the low-risk category.
- The incidence of OIRD in the three predicted risk categories is shown in **Figure 2**.
- None of the patients required naloxone during treatment with oliceridine.
- No patient experienced SpO_2 < 90% and RR < 10 bpm at the same time.
 - Based on RR < 10 bpm, patients categorized as "high-risk" on the PRODIGY score had a similar incidence of OIRD as patients with "low" or "intermediate-risk" (**Figure 2**).

LIMITATIONS

- The metrics used to measure OIRD in the PRODIGY trial were different from the ones used in the ATHENA study.
- The PRODIGY study collected continuous data whereas the ATHENA study only collected the oxygen saturation and respiration rate data at prespecified assessment time points throughout the course of the study.
- To conform with the PRODIGY metrics, we only included in the analysis patients with comorbidities of sleep apnea and/or chronic heart failure that resulted in a sample size of 103 patients.

CONCLUSIONS

- Findings from this exploratory analysis suggest that oliceridine shows a trend towards lower risk of opioid-induced respiratory depression (OIRD) in patients categorized to be at intermediate or high risk using the PRODIGY scoring tool.
- The use of oliceridine may be clinically appropriate in patients at high risk for OIRD.
- Further clinical trials are needed to confirm the findings of OIRD safety in high-risk patients.

REFERENCES

1. Small C, Laycock H. *Br J Surg*. 2020;107(2):e70-e80.
2. Gupta K, et al. *BMJ Open*. 2018;8(12):e024086.
3. Ayad S, et al. *Br J Anaesth*. 2019;123(3):378-391.
4. Khanna AK, et al. *Anesth Analg*. 2020;131(4):1012-1024.
5. DeWire SM, et al. *J Pharmacol Exp Ther*. 2013;344(3):708-717.
6. Brzezinski M, et al. *Pain Ther*. 2021;10(1):457-473.