EFFECTS OF THE ANTIDIABETIC *IMEGLIMINE* IN HYPERGLYCEMIC MICE WITH SEPTIC SHOCK

F Wagner[1]; J Vogt[1]; U Wachter [1]; S Weber [1]; B Stahl [1]; M Groeger[1]; O McCook[1]; M Georgieff[1], P Fouqueray[2], T Kuhn[2], E Calzia[1], P Radermacher[1], E Fontaine[3], K Wagner[1]

1 Anesthesiology, University Medical School, Ulm, Germany, 2 Poxel, Lyon, France, 3 LBFA, Université Joseph Fourier, Grenoble, France

Introduction:

Shock-related hyperglycemia impairs mitochondrial function and integrity [1,2], ultimately leading to apoptosis and organ failure [2,3]. Imeglimin presents as a new antidiabetic drug with anti-hyperglycemic and anti-apoptotic properties [4].Therefore we investigated its effects in hyperglycemic mice with septic shock.

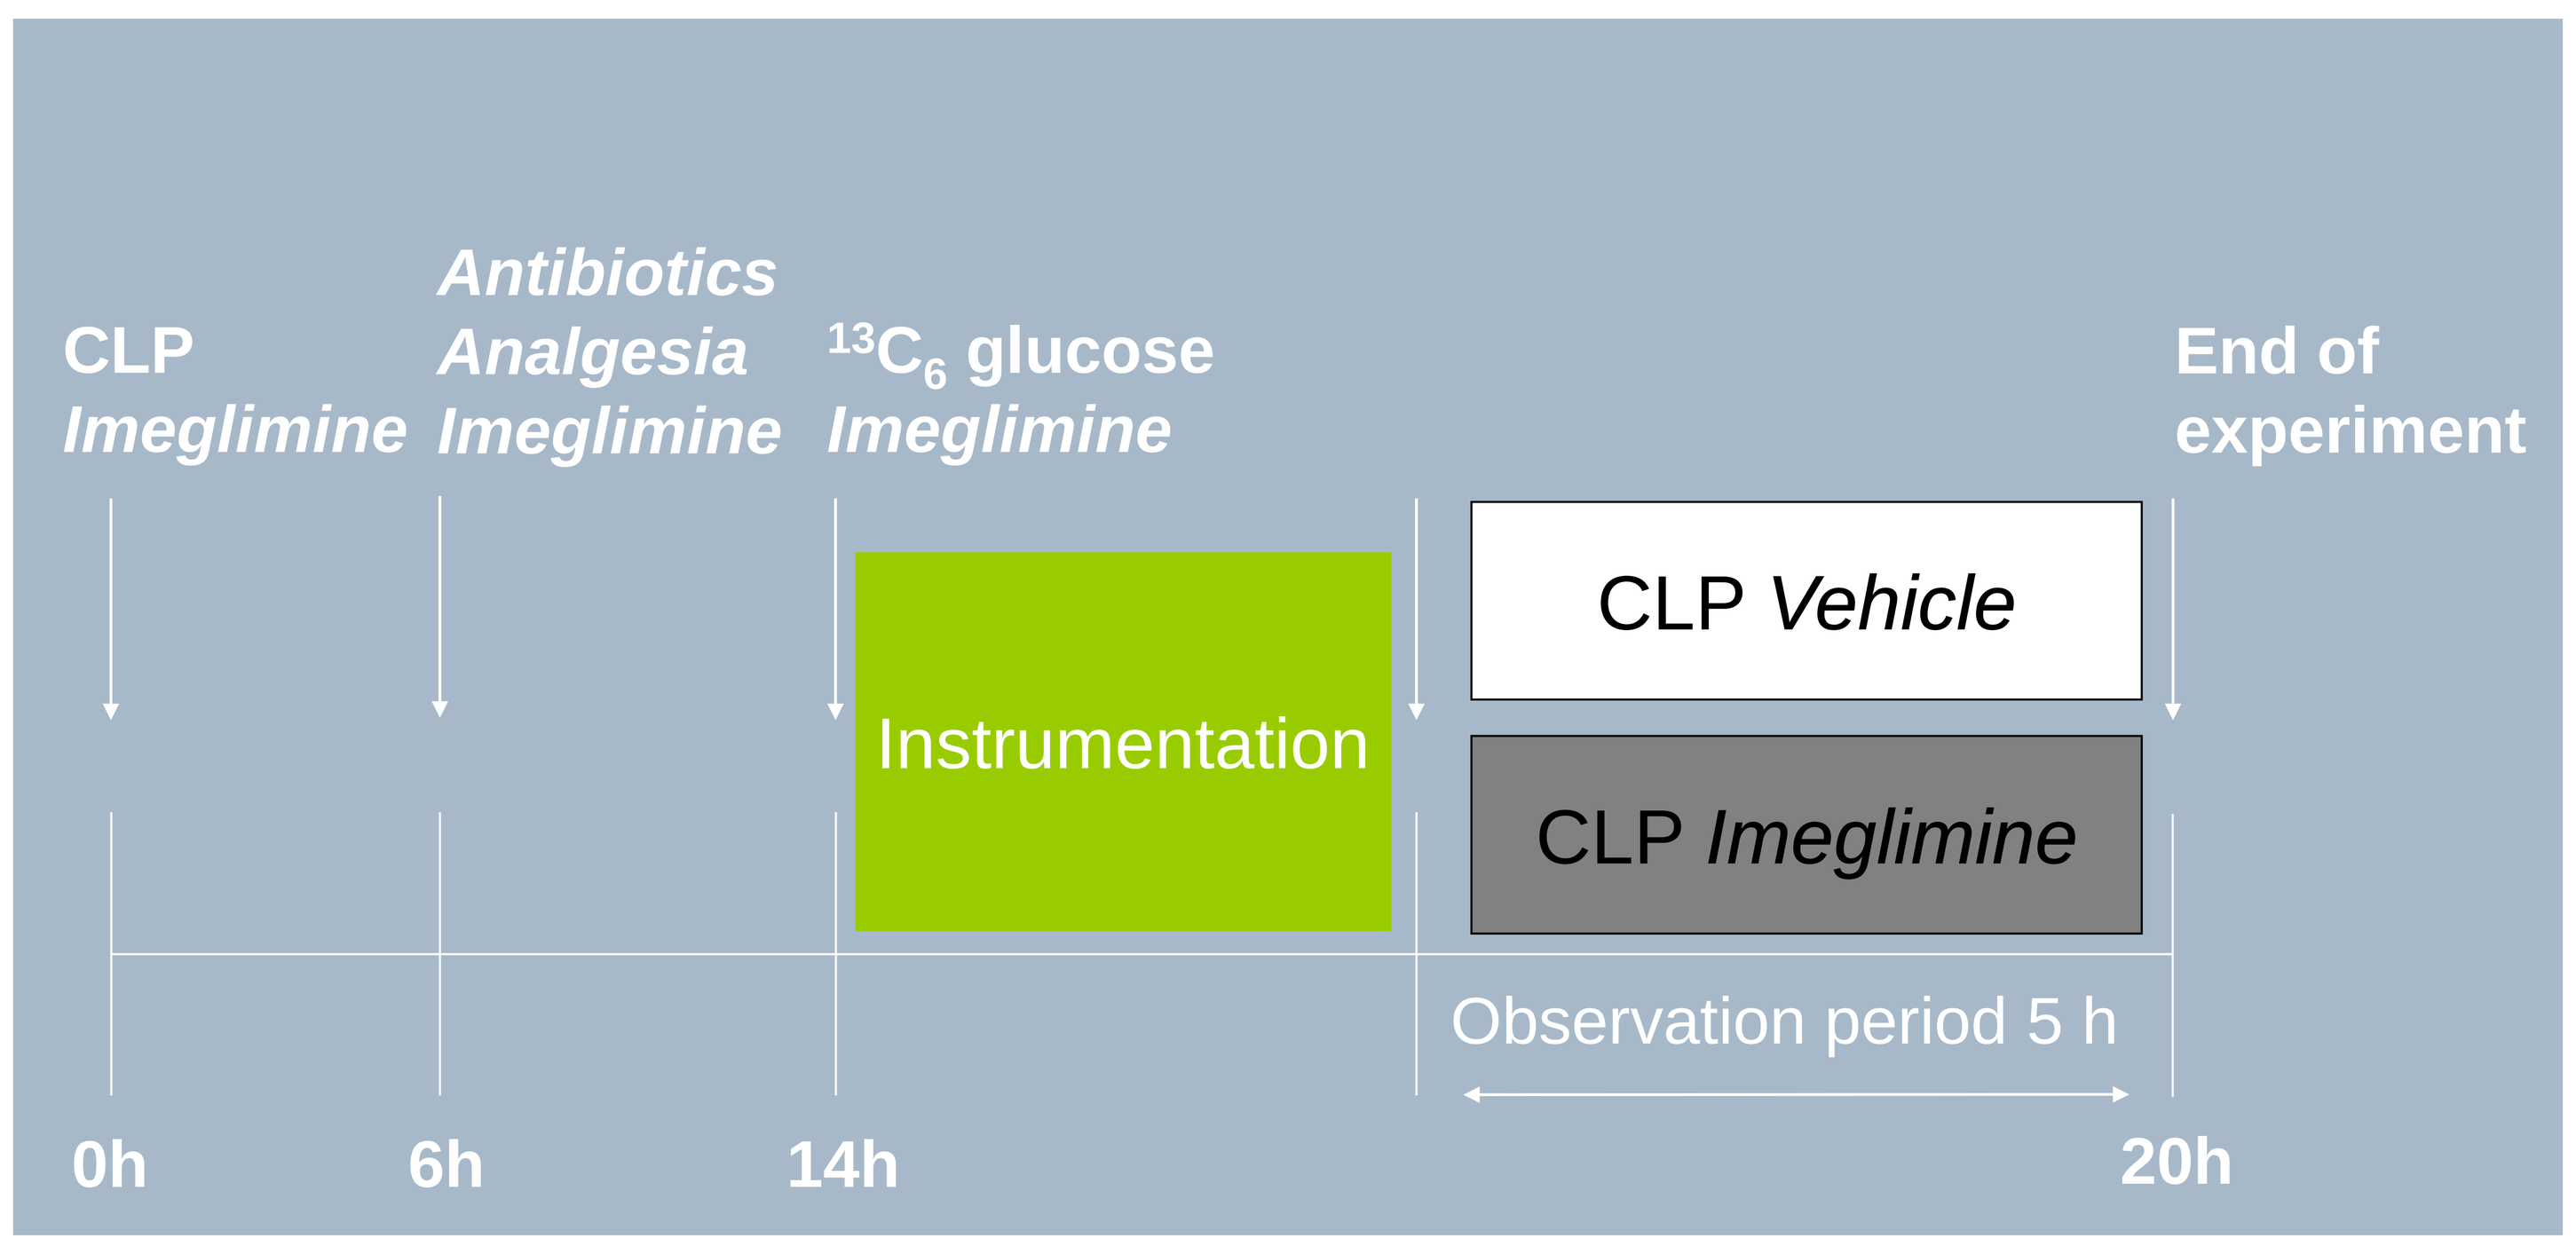
CLP
Imeglimine
Antibiotics
Analgesia
Imeglimine
$^{13}C_6$ glucose
Imeglimine
End of experiment
Instrumentation
CLP Vehicle
CLP Imeglimine
Observation period 5 h
0h
6h
14h
20h

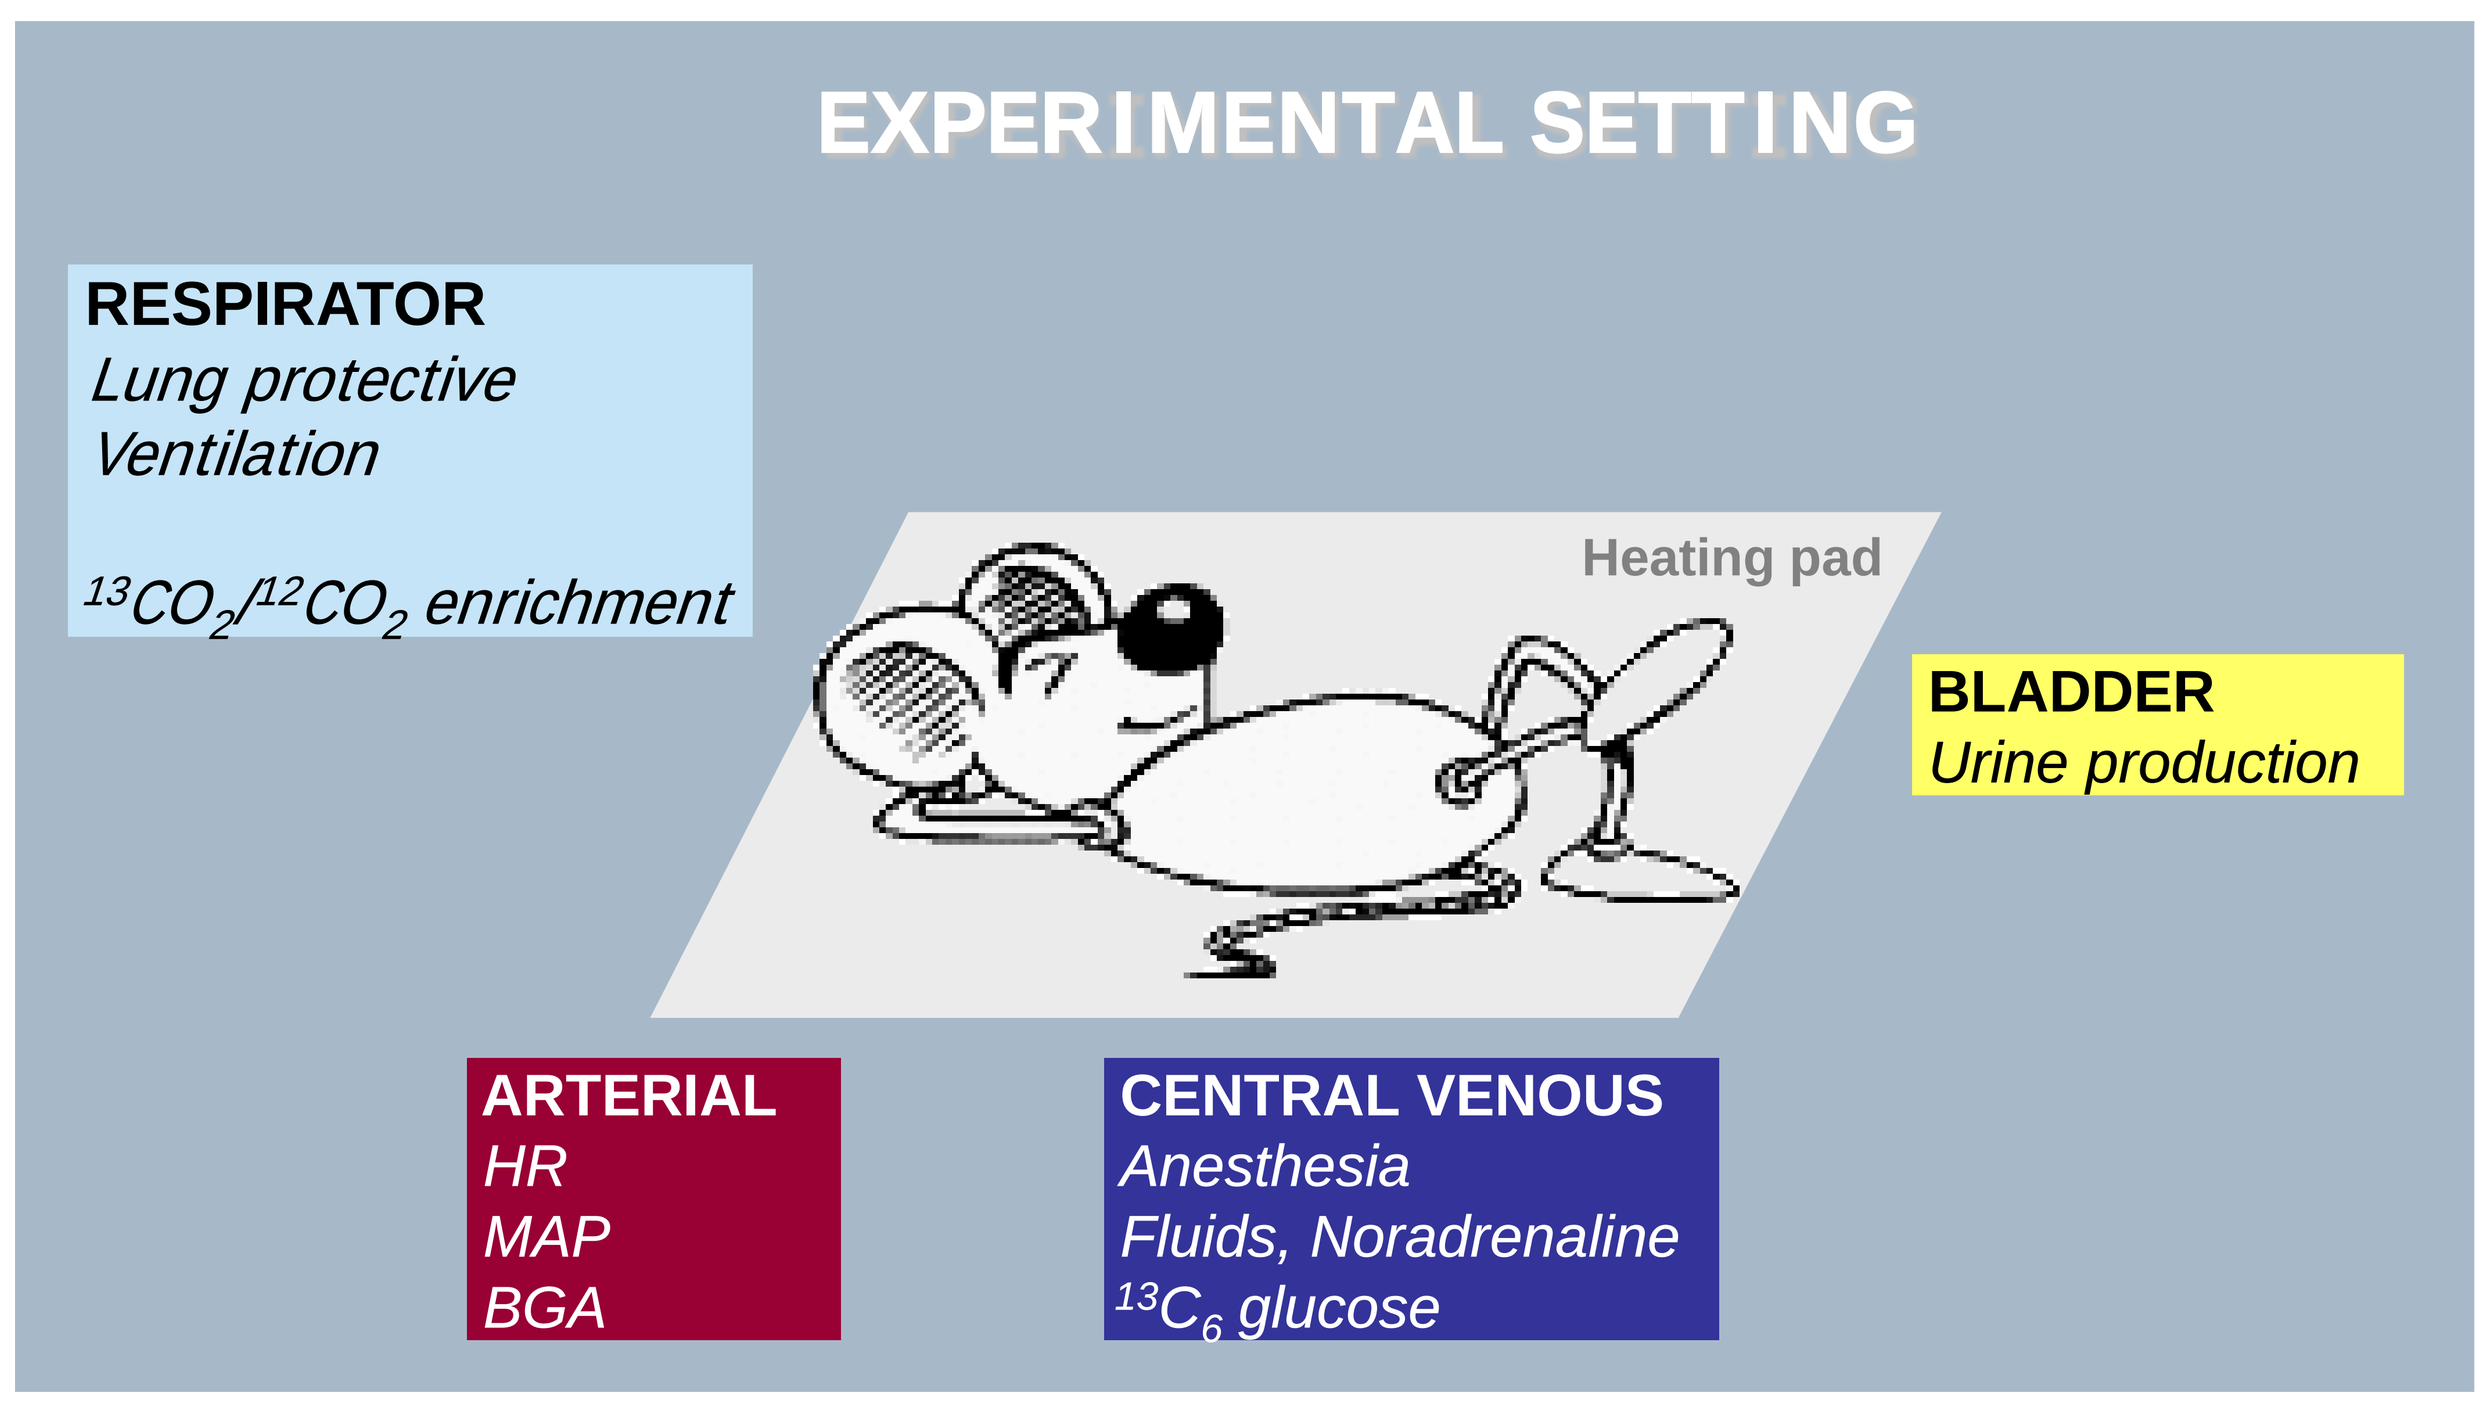
EXPERIMENTAL SETTING
RESPIRATOR
Lung protective Ventilation
$^{13}CO_2/^{12}CO_2$ enrichment
Heating pad
BLADDER
Urine production
ARTERIAL
HR
MAP
BGA
CENTRAL VENOUS
Anesthesia
Fluids, Noradrenaline
$^{13}C_6$ glucose

Methods:

Immediately and 6 hours after cecal-ligation and puncture, mice randomly received s.c. *vehicle* (n=9) or *imeglimin* (n=10; 100 µg • g^{-1}). 14 hours later animals were anesthetized, mechanically ventilated and instrumented for a consecutive 5 h observation period. After a second imeglimin bolus, colloid fluid resuscitation was performed and continuous i.v. noradrenaline were titrated to maintain normotensive and hyperdynamic hemodynamics. 2 mg • g^{-1} • h^{-1} glucose were infused to induce hyperglycemia. Glucose oxidation and gluconeogenesis were derived from blood $^{13}C_6$-glucose and mixed expiratory $^{13}CO_2/^{12}CO_2$ isotope enrichment during continuous isotope infusion. Liver mitochondrial activity was assessed using high resolution respirometry [5-7]. Liver hemoxygenase-1 (HO-1), Bcl-2–associated X protein (Bax), cleaved caspase-3 and nuclear factor-κB (NF-κB) expression by immunoblotting and EMSA, respectively. All data are median (quartiles). Statistical significance was tested using Mann-Whitney Rank sum or if data were normally distributed by t- test.

Results:

Imeglimin did neither affect noradrenaline requirements (0.15 (0.10;0.98) vs 0.13 (0.06;0.56) µg•g^{-1}, p=0.23) needed to achieve target hemodynamics nor liver macrocirculation. It decreased blood glucose levels by increasing whole body glucose oxidation, which coincided with partial restoration of gluconeogenesis. Regarding liver mitochondrial activity oxidative phosphorylation as well as maximal oxidative capacity were increased in the imeglimine group. Whereas liver HO-1 was upregulated, NF-κB as well as Bax and cleaved Caspase-3 expression were attenuated in the imeglimine treated animals.

	Vehicle		**Imeglimine**		intergroup
	Baseline	*5 h*	*Baseline*	*5 h*	
Heart rate [min^{-1}]	**490 (375;513)**	**476 (450;489)**	**435 (395;485)**	**410 (401;432)**	*n.s.*
MAP [mmHg]	**64 (61;72)**	**56 (52;58)**	**65 (65;68)**	**52 (51;56)**	*n.s.*
Flow V. portae [mL • min^{-1}]	**8 (5.9;8.4)**	**6.2 (3.3;7.3)**	**7 (6.2;7.3)**	**5 (4.5;6.1)**	*n.s.*
Liver SO_2 [%]	**67 (64;70)**	**65 (59;67)**	**70 (68;71) §**	**68(62;70)**	*§ p=0.03 vs Vehicle baseline*
pH	**7.34 (7.30;7.34)**	**7.30 (7.27;7.28)**	**7.32 (7.29;7.37)**	**7.20 (7.02;7.26)**	*n.s.*
P_aCO_2 [mmHg]	**32 (30;34)**	**35 (34;41)**	**31 (29;34)**	**38 (35;43)**	*n.s.*
P_aO_2 [mmHg]	**337 (312;366)**	**314 (292;347)**	**349 (347;357)**	**347 (336;354)**	*n.s.*
Blood lactate [mmol • L^{-1}]		**3.2 (2.5;3.5)**		**3.7 (3.1;4.7)**	*n.s.*

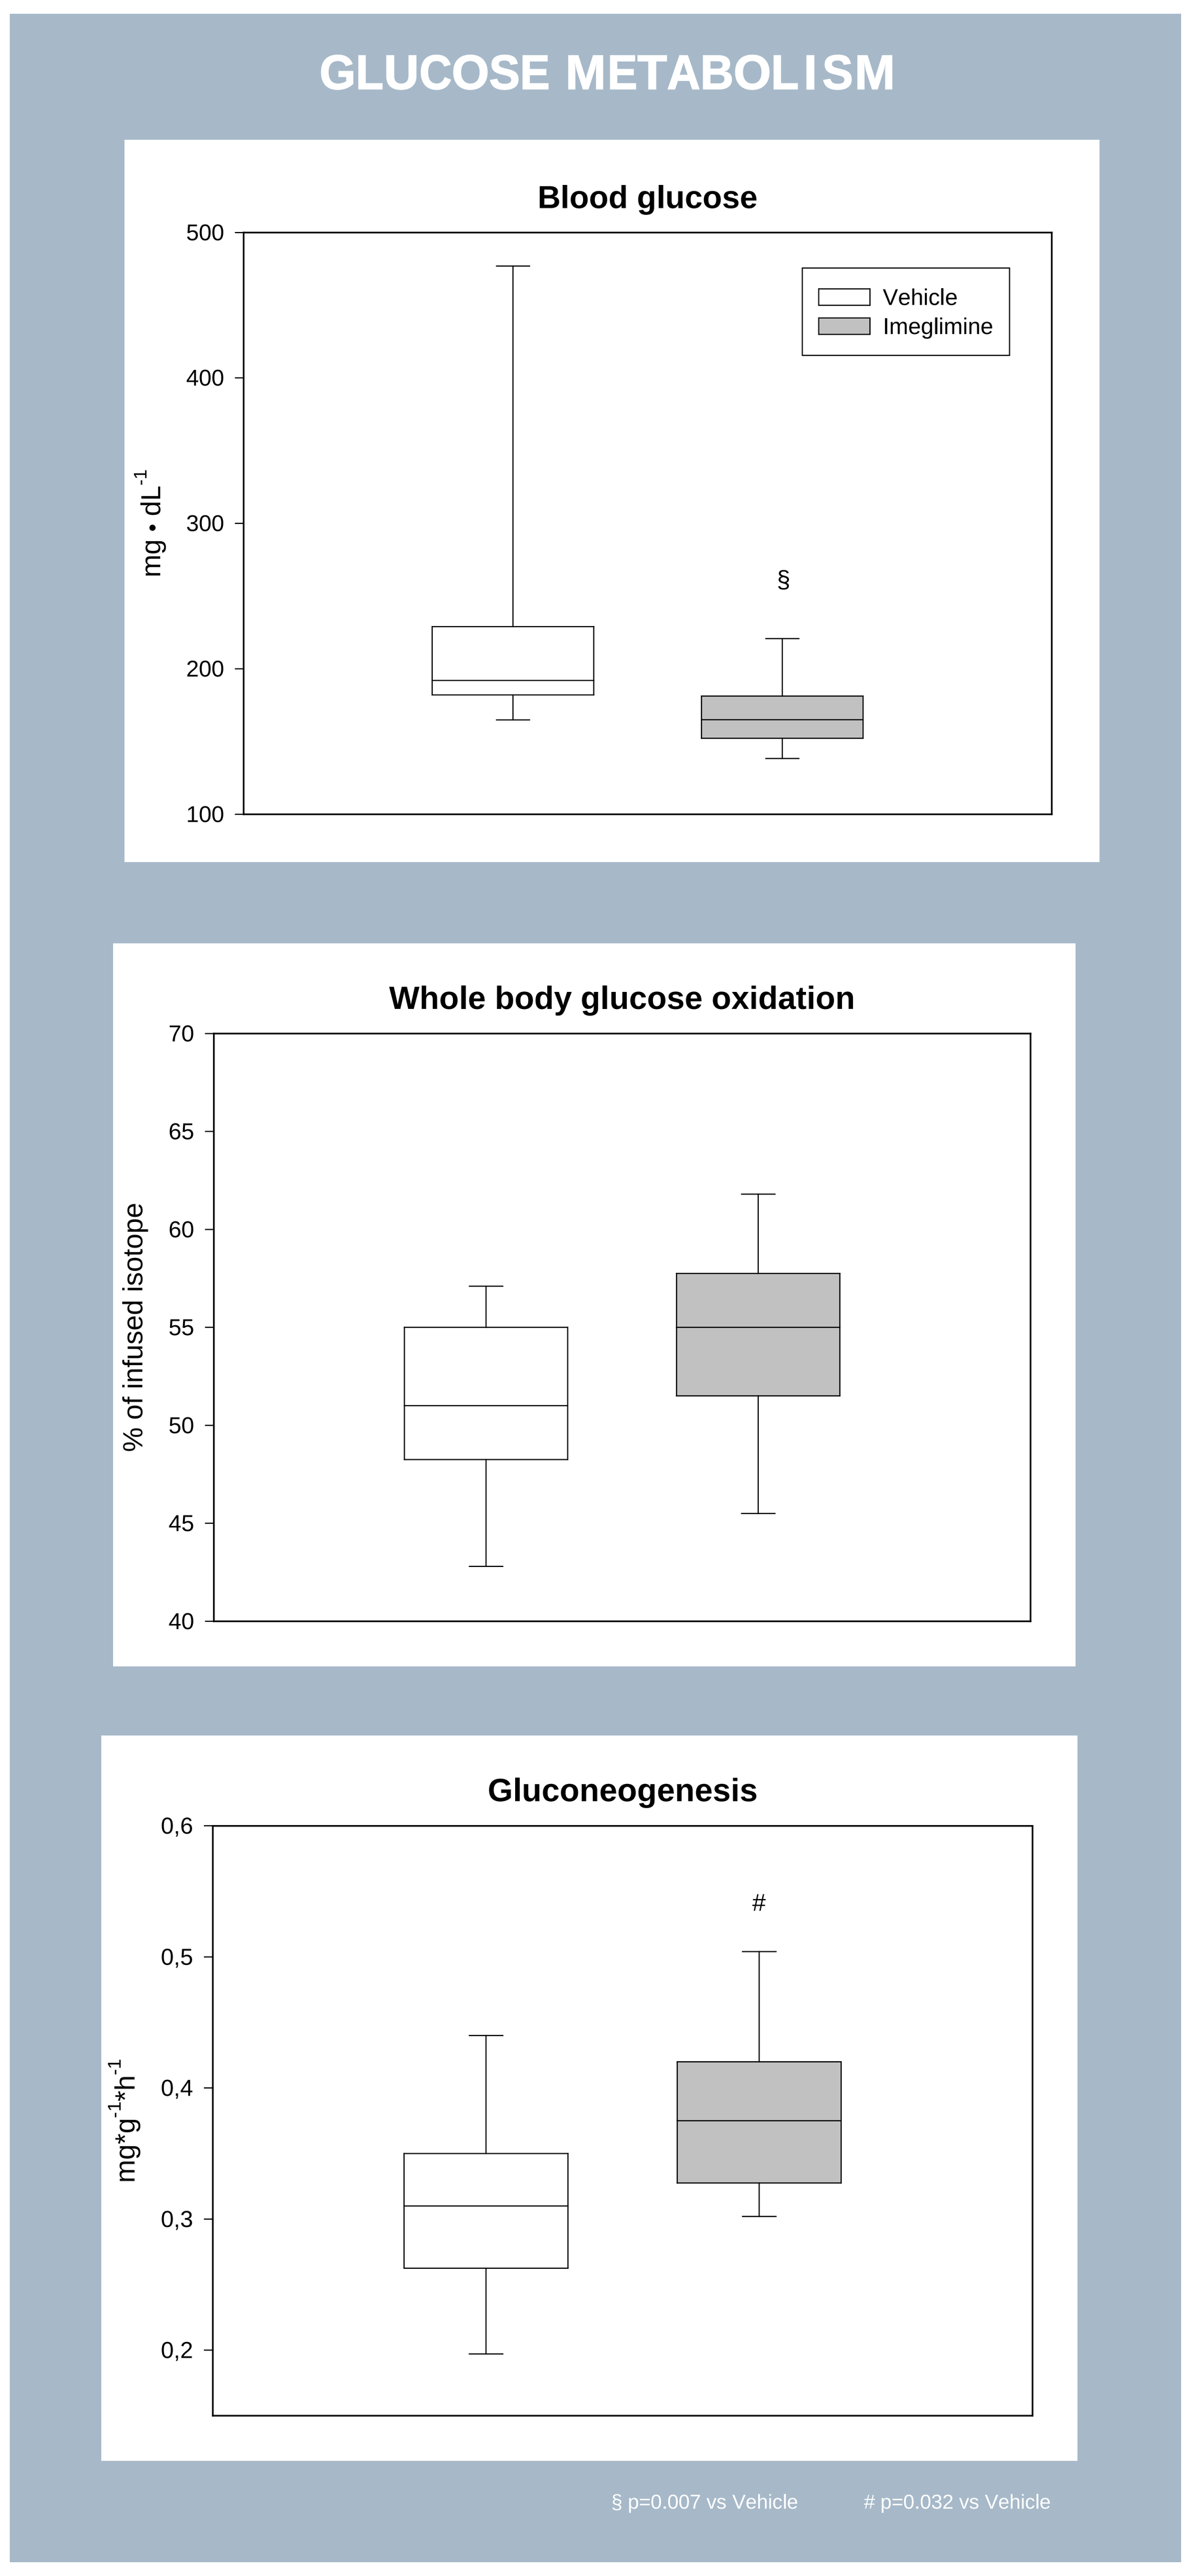
GLUCOSE METABOLISM
Blood glucose
Vehicle
Imeglimine
mg • dL^{-1}
§
Whole body glucose oxidation
% of infused isotope
Gluconeogenesis
mg*g^{-1}*h^{-1}
#
§ p=0.007 vs Vehicle
p=0.032 vs Vehicle

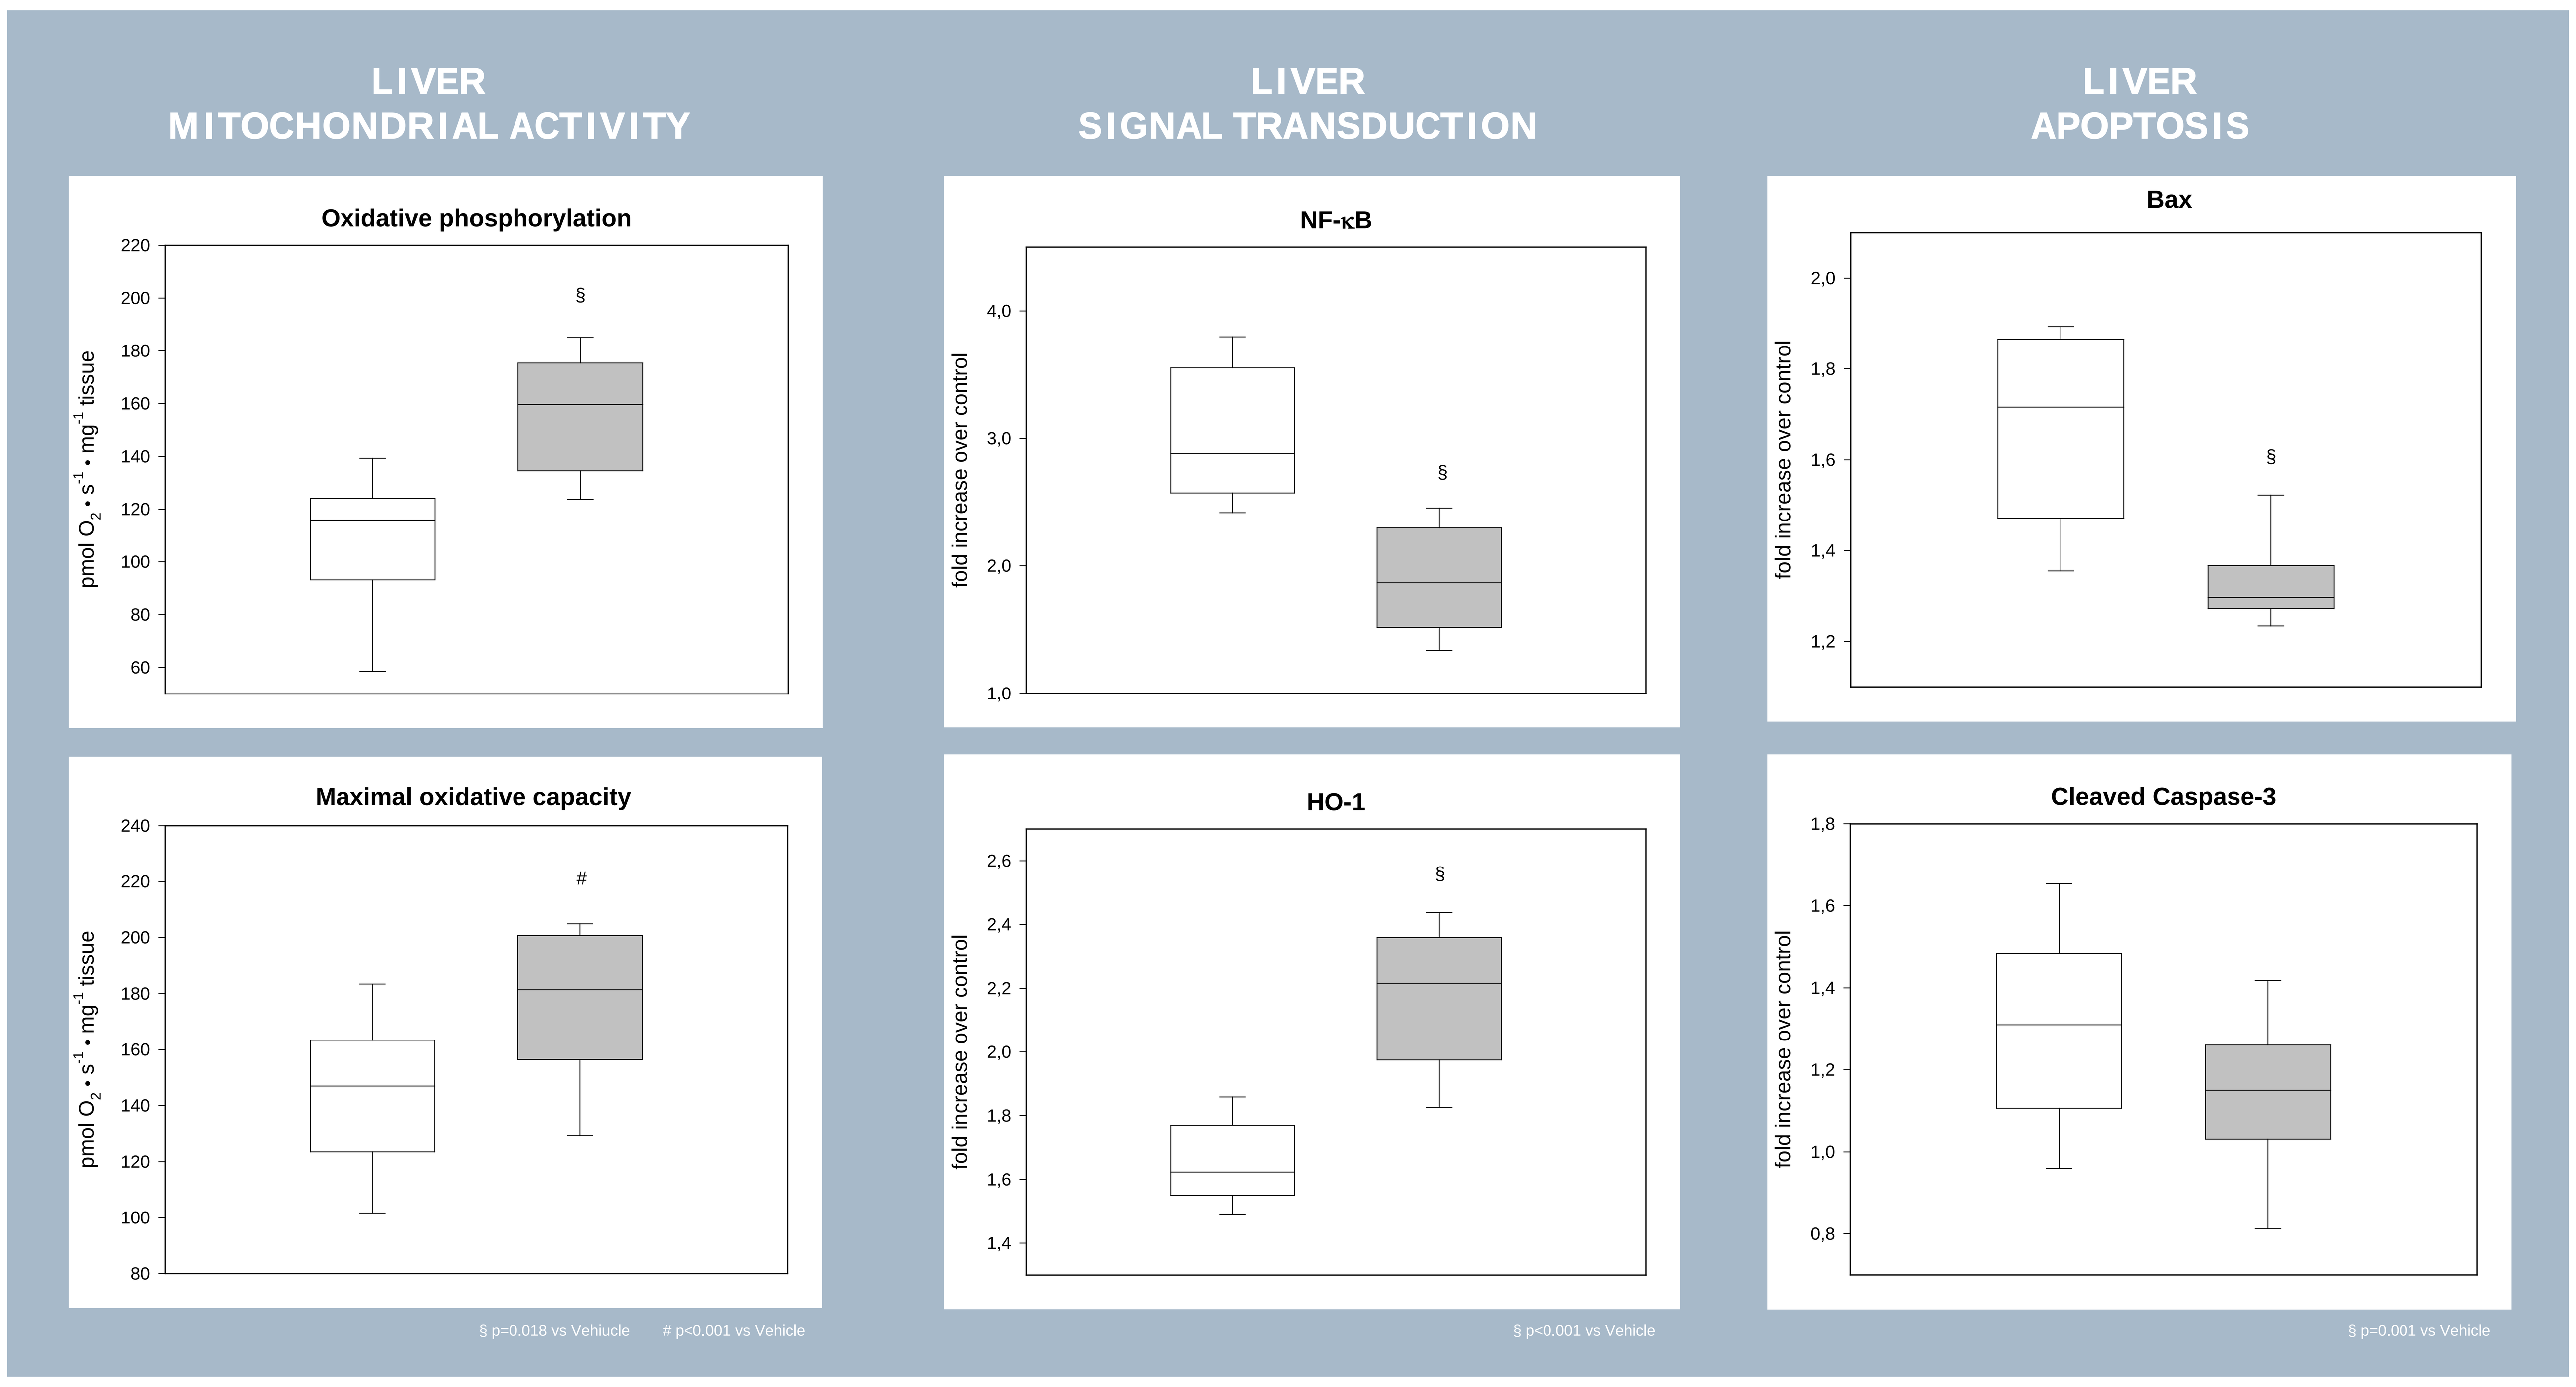
LIVER
MITOCHONDRIAL ACTIVITY
Oxidative phosphorylation
pmol O_2 • s^{-1} • mg^{-1} tissue
Maximal oxidative capacity
§ p=0.018 vs Vehiucle
p<0.001 vs Vehicle
LIVER
SIGNAL TRANSDUCTION
NF-κB
HO-1
fold increase over control
§ p<0.001 vs Vehicle
LIVER
APOPTOSIS
Bax
Cleaved Caspase-3
§ p=0.001 vs Vehicle

Conclusions:

Imeglimin improved liver mitochondrial activity as well as whole body glucose utilization and gluconeogenesis, a well-established marker of liver metabolic capacity [5,6]. This was coincided with attenuated organ injury, at least in part due to diminished NF-κB activation and inhibition of the mitochondrial apoptosis pathway.

Acknowledgements:

In memoriam of ***Xavier Leverve*** who initiated this project.
The study was supported by an unrestricted grant of Poxel.

References:

1. Vanhorebeek I, Lancet 365:53-59, 2005
2. Vanhorebeek I, CritCareMed 37:1355-64, 2009
3. Devos P, CurrOpinClinNutrMetabCare 9 :131-9, 2006
4. Fouqueray, JDiabetesMetab 2:4, 2011
5. Albuszies G, CritCareMed 33:2332-8, 2005
6. Albuszies G, IntCareMed 33:1094-1101, 2007
7. Baumgart K, CritCareMed 38:588-95, 2010

Excellence in achieving PoC