1169-P, American Diabetes Association, 5–9th June 2015, Boston, USA

Dose Ranging-Study to Determine the Optimum Dose for Imeglimin, a Novel Treatment for Type 2 Diabetes

Pascale Fouqueray, MD[1], Sebastien Bolze, PhD[1], Valdis Pirags, MD[2], Clifford J. Bailey, PhD[3], Guntram Schernthaner, MD[4], Silvio E. Inzucchi, MD[5], John E. Gerich, MD[6], Michael Roden, MD[7,8,9], Harold E. Lebovitz, MD[10]

[1]Poxel SA; [2]Paul Stradins Clinical University Hospital, Riga, Latvia; [3]Aston University, Birmingham, United Kingdom; [4]Rudolfstiftung Hospital, Vienna, Austria; [5]Yale University School of Medicine, New Haven, CT, USA; [6]University of Rochester School of Medicine, Rochester, NY, USA; [7]Institute for Clinical Diabetology, German Diabetes Center, Leibniz Institute for Diabetes Research at Heinrich Heine University Düsseldorf, Düsseldorf, Germany; [8]German Center for Diabetes Research (DZD e.V.), Partner Düsseldorf, Germany; [9]Department of Endocrinology and Diabetology, Medical Faculty, Heinrich Heine University Düsseldorf, Düsseldorf, Germany; [10]State University of New York, Health Sciences Center, Brooklyn, NY, USA

Background

- Imeglimin is the first in a new tetrahydrotriazine-containing class of oral anti-diabetic agents, the glimins
- Imeglimin targets mitochondrial bioenergetics and improves abnormal features of mitochondrial function leading to an increase in fatty oxidation and a decrease in ROS overproduction[1]
- Imeglimin has been shown to induce glucose-stimulated insulin secretion by improving beta cell glucose sensitivity in patients with type 2 diabetes[2] and to improve insulin sensitivity in a rodent diabetic model allowing normalization of glucose tolerance[1]
- The efficacy of imeglimin has been demonstrated in T2D patients, both as mono and add-on (metformin and sitagliptin) therapies with no safety signals to date (600 subjects)[3–5]
- Imeglimin may therefore be a novel antihyperglycemic agent with a unique mechanism of action that complements those of drugs that act on insulin-resistant organs or on insulin secretion

Objectives

- This Phase IIb study assessed the dose-response of imeglimin at 4 doses compared to placebo in male and female subjects with T2D after 24 weeks of treatment, using A1C reduction from baseline as the primary evaluation criterion

Research Design and Methods

- This was a 24 week, multicenter, double-blind, placebo-controlled, randomized study in subjects with T2D, in 5 parallel groups: 4 groups of imeglimin doses (500, 1000, 1500, or 2000 mg twice daily) and 1 placebo group
- Subjects were screened within 3 weeks, followed by a 3-week placebo run-in period for treatment-naïve subjects, or 6-week placebo wash-out/run-in period for subjects previously treated with an oral antidiabetic monotherapy

Figure 1 – Imeglimin dose-ranging study design

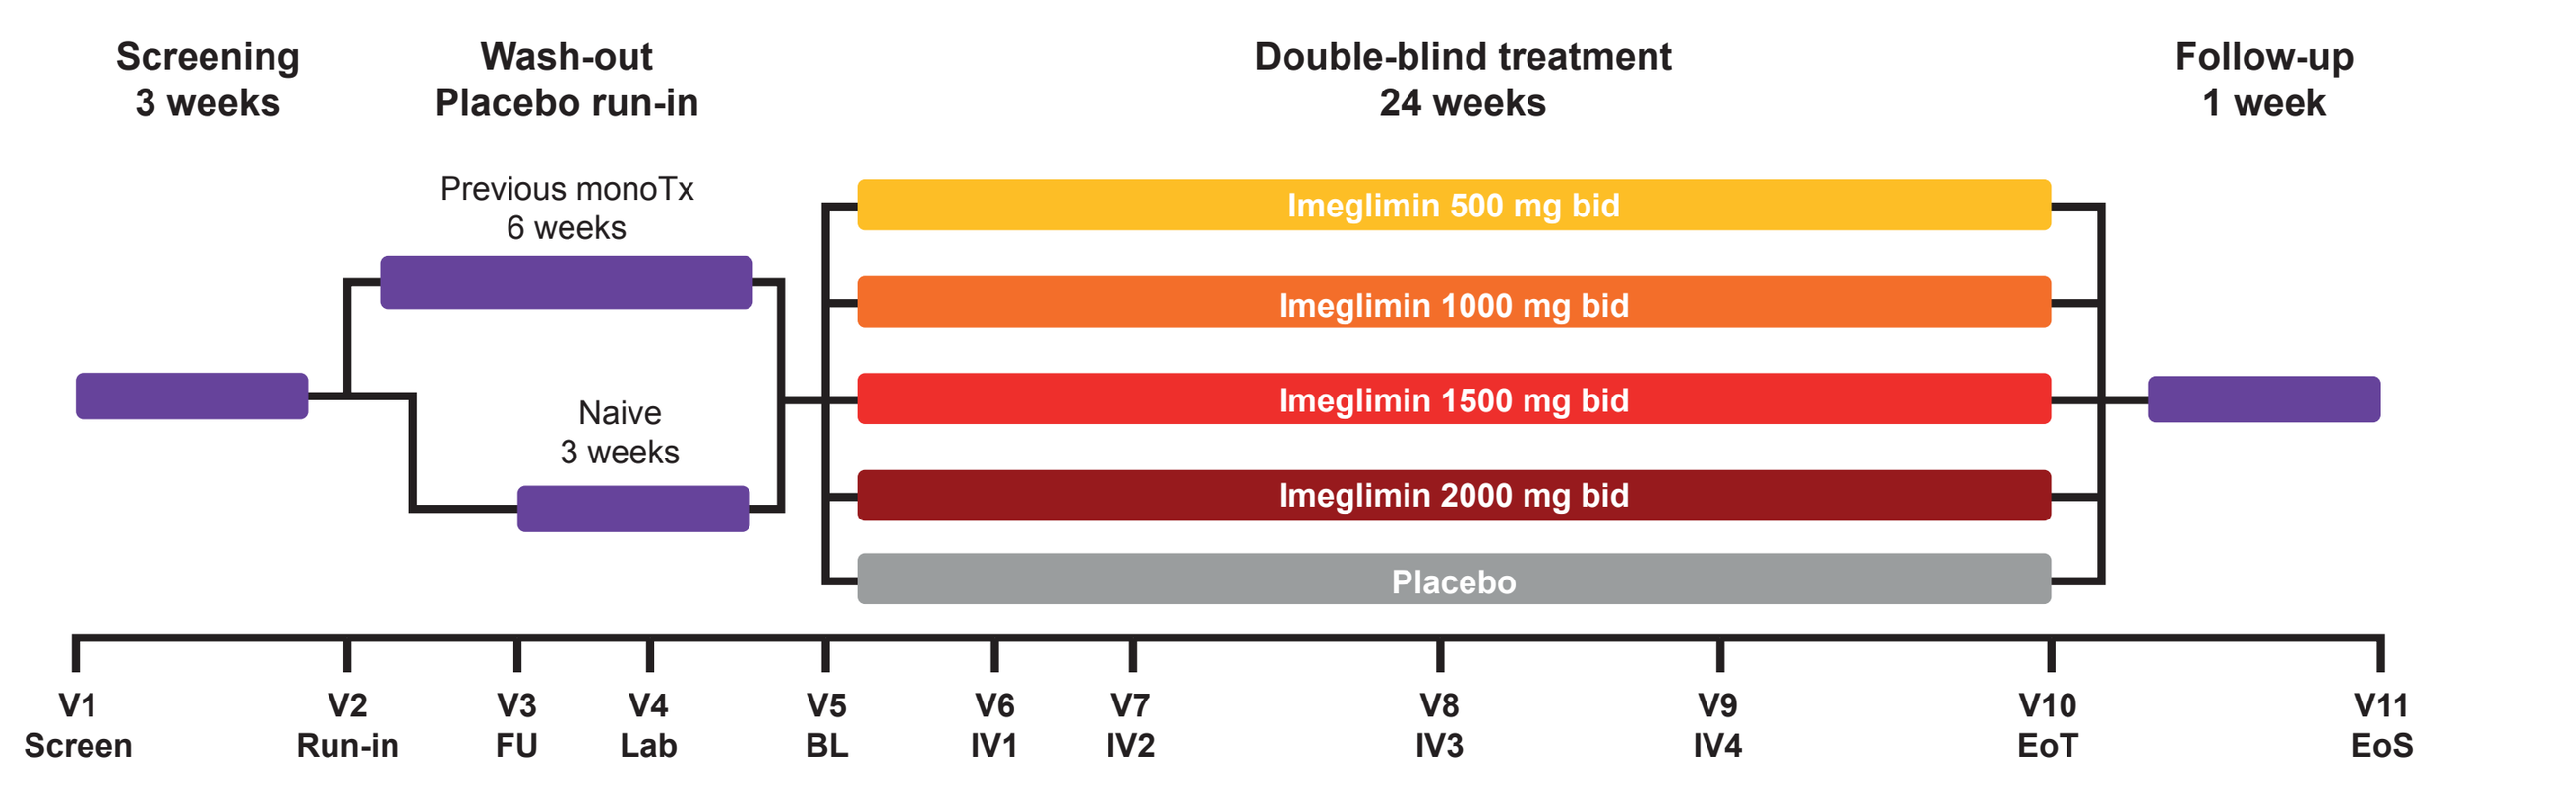
Screening
3 weeks
Wash-out
Placebo run-in
Double-blind treatment
24 weeks
Follow-up
1 week
Previous monoTx
6 weeks
Naive
3 weeks
Imeglimin 500 mg bid
Imeglimin 1000 mg bid
Imeglimin 1500 mg bid
Imeglimin 2000 mg bid
Placebo
V1 Screen
V2 Run-in
V3 FU
V4 Lab
V5 BL
V6 IV1
V7 IV2
V8 IV3
V9 IV4
V10 EoT
V11 EoS

Bid, twice daily; BL, baseline; EoS, end-of-study; EoT, end-of-treatment; FU, follow-up; IV, intermediate visit; Lab, laboratory; monoTx, monotherapy; Screen, screening; V, visit

- Subjects were then randomized 1:1:1:1:1 to receive either 1 of the 4 doses of imeglimin or placebo in a parallel group and double-blind manner for 24 weeks, followed by a 1-week follow up period with placebo
- The primary efficacy endpoint of the study was the placebo-adjusted dose-dependent reduction in A1C from baseline to week 24
- Secondary end points included changes from baseline in FPG, % A1C responders (≤7%), and non-glycemic parameters
- The safety and tolerability of 24-week treatment with each imeglimin dose versus placebo were assessed, with adverse events categorized by severity

Results - Patients

- In total, 382 subjects were randomized for the study (Table 1)

Table 1: Subject disposition

	Imeglimin 500 mg bid	Imeglimin 1000 mg bid	Imeglimin 1500 mg bid	Imeglimin 2000 mg bid	Placebo	Total
Randomized	**74**	**79**	**74**	**74**	**81**	**382**
Naïve	18 (24.3)	16 (20.3)	17 (23.0)	20 (27.0)	23 (28.4)	94 (24.6)
Previously treated	56 (75.7)	63 (79.7)	57 (77.0)	54 (73.0)	58 (71.6)	288 (75.4)
Safety	**74 (100)**	**79 (100)**	**74 (100)**	**74 (100)**	**81 (100)**	**382 (100)**
ITT	**72 (97.3)**	**77 (97.5)**	**69 (93.2)**	**69 (93.2)**	**80 (98.8)**	**367 (96.1)**
PP	**62 (83.8)**	**65 (82.3)**	**60 (81.1)**	**61 (82.4)**	**63 (77.8)**	**311 (81.4)**
Discontinuation	**11 (14.9)**	**13 (16.5)**	**14 (18.9)**	**13 (17.6)**	**16 (19.8)**	**67 (17.5)**
Withdrawal of consent	4 (5.4)	4 (5.1)	8 (10.8)	5 (6.8)	7 (8.6)	28 (7.3)
Rescue	5 (6.8)	6 (7.6)	0	4 (5.4)	6 (7.4)	21 (5.5)
Adverse event	0	1 (1.3)	2 (2.7)	3 (4.1)	0	6 (1.6)
Protocol deviation	1 (1.4)	2 (2.5)	1 (1.4)	1 (1.4)	0	5 (1.3)
Other	0	0	2 (2.7)	0	1 (1.2)	3 (0.8)
Non permitted medication	0	0	1 (1.4)	0	1 (1.2)	2 (0.5)
Non compliance	1 (1.4)	0	0	0	0	1 (0.3)
Exclusion criteria	0	0	0	0	1 (1.2)	1 (0.3)

Data are n (%)

ITT, intent-to-treat; PP, per protocol

- Baseline characteristics were balanced between all treatment groups (Table 2)

Table 2: Demographics and baseline characteristics

	Imeglimin 500 mg bid	Imeglimin 1000 mg bid	Imeglimin 1500 mg bid	Imeglimin 2000 mg bid	Placebo
Age, years (SD)	58.1 (10.6)	58.5 (8.9)	58.1 (8.5)	59.0 (8.8)	58.2 (9.3)
Gender, female, n (%)	40 (54.1)	54 (68.4)	45 (60.8)	45 (60.8)	44 (54.3)
BMI, kg/m² (SD)	31.8 (4.5)	31.4 (4.4)	32.0 (4.5)	31.0 (4.5)	30.5 (4.2)
Duration of diabetes					
Mean (SD)	5.2 (4.4)	5.4 (4.1)	6.0 (5.5)	5.7 (5.1)	5.0 (4.1)
Median	4.2	5.1	5.5	4.7	4.1
Min/max	0.3/19.2	0.0/14.2	0.0/28.8	0.1/33.3	0.0/16.2
A1C, % (SD)	7.95 (0.69)	8.09 (0.77)	7.89 (0.59)	8.04 (0.74)	7.76 (0.62)
FPG, mmol/L (SD)	9.92 (2.80)	10.29 (2.14)	9.99 (2.18)	9.76 (2.40)	9.63 (2.12)
HOMA IR (SD)	7.3 (4.7)	8.1 (5.9)	8.1 (6.5)	7.6 (5.2)	7.0 (4.9)

BMI, body mass index; HOMA IR, homeostasis model assessment of insulin resistance

Efficacy

- Placebo-adjusted reductions in A1C from baseline were observed with all doses of imeglimin, with significant reductions observed in the 1500 mg bid (–0.63%, *P*<0.001) and 2000 mg bid groups (–0.50%, *P*=0.002) (Figure 2A)
- Maximal efficacy for all doses of imeglimin was obtained after 18 weeks of treatment (Figure 2B)

Figure 2 – Placebo-adjusted reduction in A1C at 24 weeks (A) and over time (B)

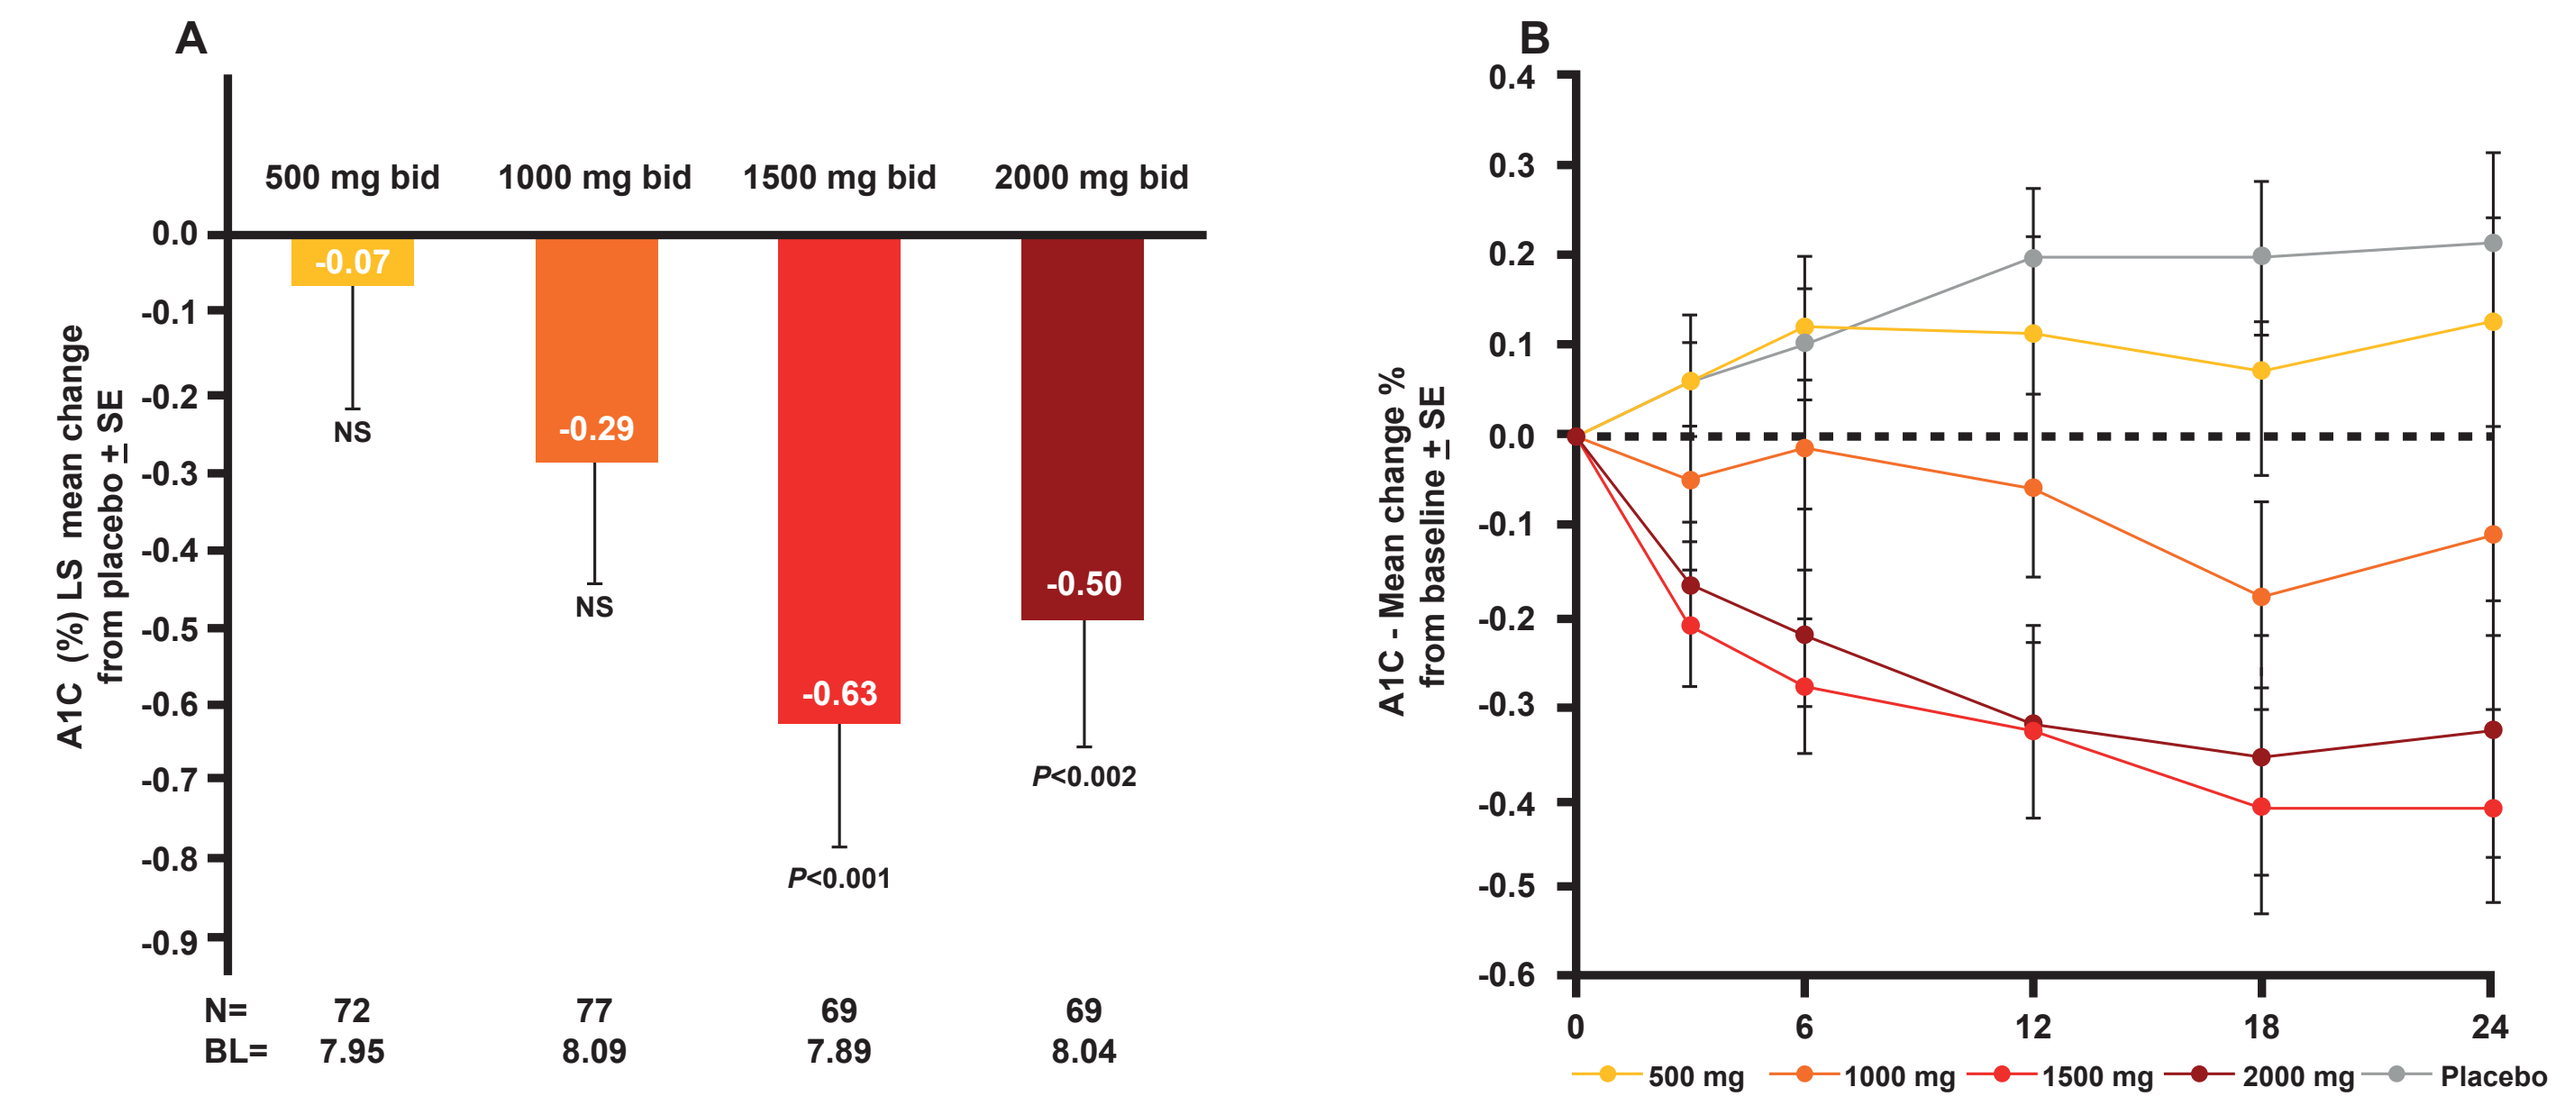
A
500 mg bid
1000 mg bid
1500 mg bid
2000 mg bid
A1C (%) LS mean change from placebo ± SE
-0.07
NS
-0.29
NS
-0.63
P<0.001
-0.50
P<0.002
N= 72 77 69 69
BL= 7.95 8.09 7.89 8.04
B
A1C - Mean change % from baseline ± SE
0 6 12 18 24
500 mg
1000 mg
1500 mg
2000 mg
Placebo

One subject in the 2000 mg bid group was excluded from the A1C and all other efficacy analyses due to blood sample error at Visit 10

- Significant placebo-adjusted reductions in FPG were also observed for the 1500 mg bid (–1.25 mmol/L, *P*=0.001) and 2000 mg bid groups (–0.81 mmol/L, *P*=0.029)
- A significantly greater proportion of patients achieved an A1C ≤7% at week 24 with imeglimin 1500 mg bid (33.3%, *P*=0.005), compared with placebo (Figure 3)
- No subjects in the imeglimin 1500 mg bid group required rescue treatment up to Week 24 (Figure 4)

Figure 3 – Proportion of patients achieving and A1C ≤7% at Week 24

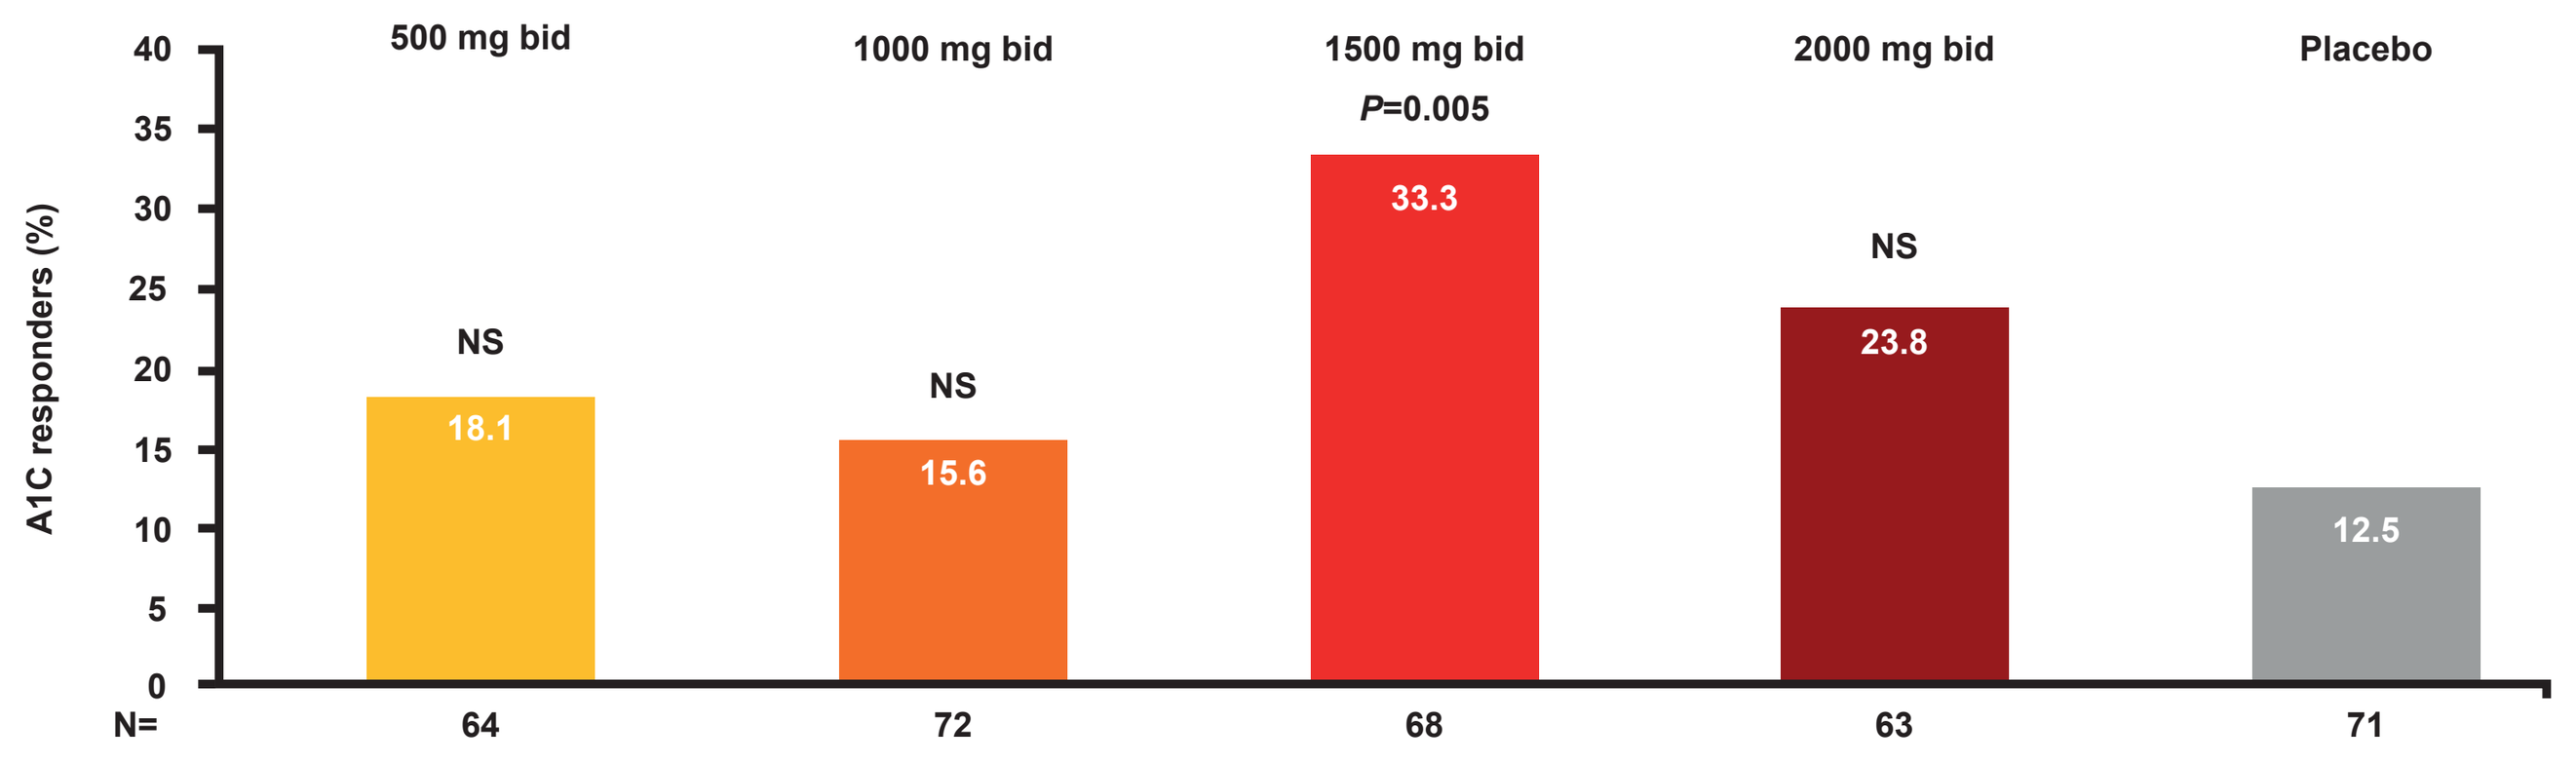
A1C responders (%)
500 mg bid
1000 mg bid
1500 mg bid
2000 mg bid
Placebo
NS
18.1
NS
15.6
P=0.005
33.3
NS
23.8
12.5
N= 64 72 68 63 71

Figure 4 – Proportion of patients requiring rescue therapy up to 24 weeks

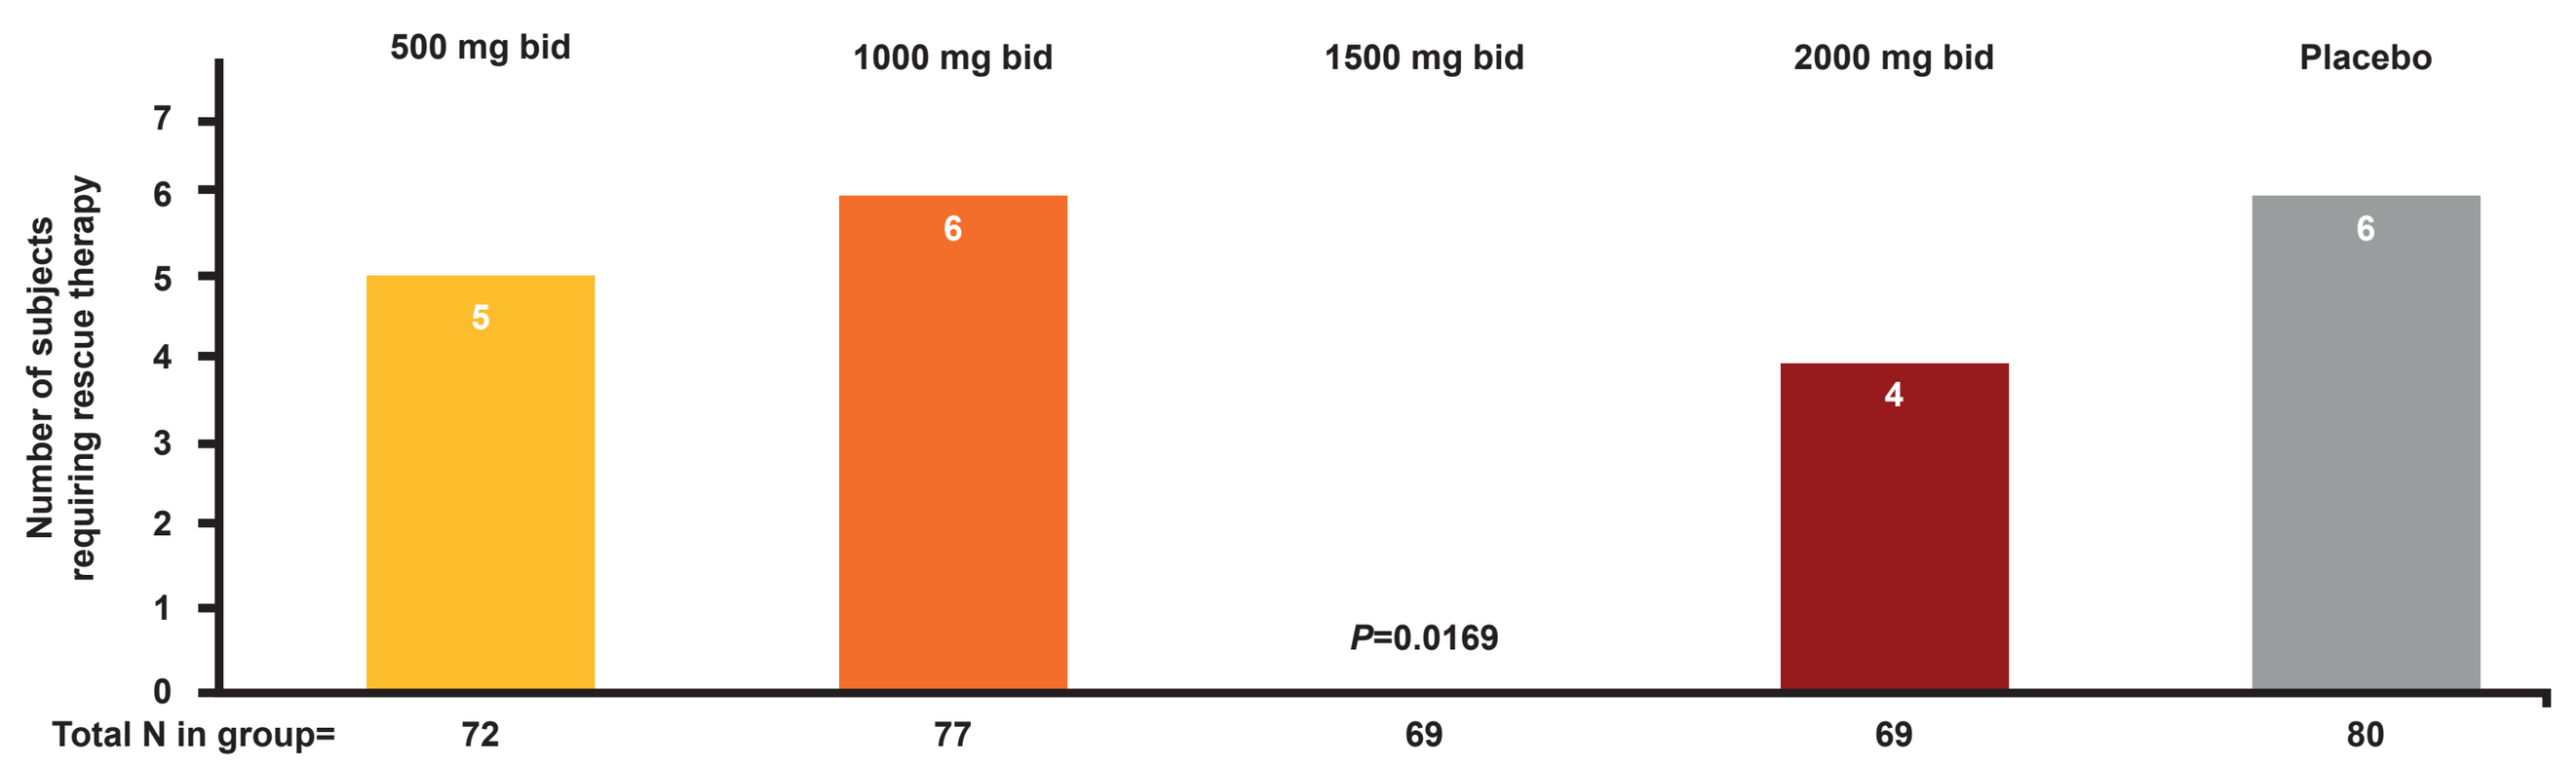
Number of subjects requiring rescue therapy
500 mg bid
1000 mg bid
1500 mg bid
2000 mg bid
Placebo
5
6
P=0.0169
4
6
Total N in group= 72 77 69 69 80

- Imeglimin (all doses) had no measurable effects on fasting insulin and C-peptide concentrations compared with placebo
- Imeglimin (all doses) had neutral effects on other cardiovascular risk factors such as body weight and lipids compared with placebo

Safety

- All doses of imeglimin well tolerated with a comparable safety profile to placebo (Table 3)
- The most commonly reported related treatment-emergent adverse events in the imeglimin 2000 mg bid group were gastrointestinal in nature (7 subjects, 17 events); 3 subjects in this group discontinued treatment due to adverse events
- There were no reported serious treatment-emergent adverse events related to imeglimin or placebo; no episodes of hypoglycemia were reported in the study

Table 3: Safety and tolerability overview

Adverse Event, N (%) - E	Imeglimin 500 mg bid n=81	Imeglimin 1000 mg bid n=79	Imeglimin 1500 mg bid n=74	Imeglimin 2000 mg bid n=74	Placebo n=81
Any TEAE	30 (40.5) – 69	41 (51.9) – 74	30 (40.5) – 62	41 (55.4) - 72	36 (44.4) – 76
Any serious TEAE	-	2 (2.5) – 2	1 (1,4) – 1	-	1 (1.2) – 2
Any serious related TEAE	-	-	-	-	-
Any TEAE leading to discontinuation	-	1 (1.3) – 1	2 (2.7) – 9	3 (4.1) – 7	-
Any related TEAE	6 (8.1) – 9	13 (16.5) – 15	5 (6.8) – 12	13 (17.6) – 25	6 (7.4) – 12
Gastrointestinal	**2 (2.7) – 3**	**1 (1.3) – 1**	**3 (4.1) – 7**	**7 (9.5) – 17**	**-**
Abdominal pain	-	-	2 (2.7) – 3	2 (2.7) – 4	-
Constipation	1 (1.4) – 1	-	-	-	-
Diarrhea	1 (1.4) – 1	-	1 (1.4) – 1	4 (5.4) – 7	-
Dyspepsia	-	1 (1.4) – 1	-	1 (1.4) – 1	-
Nausea	1 (1.4) – 1	-	2 (2.7) – 2	3 (4.1) – 4	-
Vomiting	-	-	1 (1.4) – 1	1 (1.4) – 1	-
Metabolism-nutrition	**3 (4.1) – 4**	**9 (11.4) – 10**	**-**	**7 (9.5) – 7**	**5 (6.2) – 8**
Hyperglycemia	2 (2.7) – 3	9 (11.4) – 10	-	7 (9.5) – 7	4 (4.9) – 7
Increased appetite	1 (1.4) – 1	-	-	-	1 (1.2) – 1
Nervous system	**-**	**1 (1.3) – 1**	**3 (4.1) – 3**	**1 (1.4) – 1**	**1 (1.2) – 1**
Dysgueusia	-	-	1 (1.4) – 1	-	-
Headache	-	1 (1.3) – 1	2 (2.7) – 2	1 (1.4) – 1	-
Somnolence	-	-	-	-	1 (1.2) – 1
Investigations	**2 (2.7) – 2**	**3 (3.8) – 3**	**1 (1.4) – 1**	**-**	**1 (1.2) – 1**
Lactate	-	3 (3.8) – 3	1 (1.4) – 1	-	-
ASAT	1 (1.4) – 1	-	-	-	-
Myelocyte	-	-	-	-	1 (1.2) – 1
Weight increase	1 (1.4) – 1	-	-	-	-
General disorders	-	-	**1 (1.4) – 1**	**-**	**1 (1.2) – 2**
Asthenia/thirst	-	-	1 (1.4) – 1	**-**	1 (1.2) – 2
Any MACE	-	-	**-**	**-**	-

ASAT, aspartate aminotransferase; E, number of events; MACE, major adverse cardiovascular events; N, number of subjects exposed; TEAE, treatment-emergent adverse event

Discussion

- Significant reductions in A1C, compared with placebo, were observed with the 1500 mg bid and 2000 mg bid doses of imeglimin, with maximal efficacy observed at 18 weeks
- The results of this study, along with previously published data[4,5], suggest that imeglimin significantly improves glycemic control with no hypoglycemia, and no weight gain
- Previous data from the Phase II program have demonstrated similar additive effects when imeglimin is combined with either metformin or sitagliptin, therefore making imeglimin a new complementary treatment option for patients with T2D

Conclusion

- This Phase IIb dose ranging study has demonstrated that imeglimin 1500 mg bid provides efficacious glycemic control, is well tolerated, with no increased risk of hypoglycemia and neutral effects on body weight, and is therefore the optimum dose for Phase III development

References

1. Vial G, et al. Diabetes 2015; 64:1–11
2. Pacini G et al, Diabetes, Obesity and Metabolism 2015; 17:541-545
3. Pirags V, et al. Diabetes, Obesity and Metabolism 2012;14:852–858
4. Fouqueray P, et al. Diabetes Care 2013;36:565–568
5. Fouqueray P, et al. Diabetes Care 2014;10:1935–5548

ANNO 1910
PAULS STRADINS
CLINICAL UNIVERSITY HOSPITAL

poxel
Excellence in achieving PoC

Pascale Fouqueray, 200 avenue Jean Jaurès F-69007 Lyon,
pascale.fouqueray@poxelpharma.com