1081-P, American Diabetes Association, 10th–14th June, New Orleans, LA, USA

Imeglimin Increases Insulin Secretion in Response to Glucose as a Unique Mechanism of Action Depending on NAD Synthesis

Sophie Hallakou-Bozec[1], Sebastien Bolze[1], Micheline Kergoat[2], Michael Roden[3–5]

[1]Poxel SA, 259-261 avenue Jean Jaurès, 69007 Lyon, France; [2]Metabrain Research, 4 avenue du Président François Mitterrand, 91380 Chilly-Mazarin, France; [3]Institute for Clinical Diabetology, German Diabetes Center, Leibnitz Center for Diabetes Research, Auf'm Hennekamp 65, 40225 Düsseldorf, Germany; [4]German Center for Diabetes Research (DZD e.V.), Partner Düsseldorf, Germany; [5]Department of Endocrinology and Diabetology, Medical Faculty, Heinrich Heine University Düsseldorf, Düsseldorf, Germany

Background

- Imeglimin is the first in a new tetrahydrotriazine-containing class of oral glucose-lowering agents, the glimins[1]
- Imeglimin's mechanism of action (MOA) leads to a potentiation of glucose-stimulated insulin secretion (GSIS), as previously demonstrated *in vivo* in animal models of diabetes[2] and in patients with type 2 diabetes[3]
- The unique MOA of Imeglimin involves the regulation of mitochondrial bioenergetics, as observed in a high-fat, high-sucrose mouse model[2]
- Nicotinamide adenine dinucleotide (NAD) is pivotal to mitochondrial function and the effects of Imeglimin on this pathway could contribute to the improvement of glucose homeostasis in patients with type 2 diabetes

Objectives

- The aim of this study is to identify the pathways involved in Imeglimin glucose-dependent insulin secretion

Research Methods

- Islets were isolated by collagenase method from Goto-Kakizaki (GK) rats euthanized on the day of the experiment

Effect of Imeglimin on insulin secretion from GK rat islets

- Islets were incubated in 2.8 mM glucose or 16.7 mM glucose, and in the absence or presence of Imeglimin (25, 50, and 100 µM) or glucagon-like peptide-1 (GLP-1; 0.1 µM) for 20 min. Insulin levels were measured by enzyme-linked immunosorbent assay (ELISA) (Alpco 80-INSRTU-E01)

Effect of Imeglimin on NAD content of GK rat islets

- Islets were incubated in the presence of 2.8 mM glucose or 16.7 mM glucose ± Imeglimin (100 µM), or ± nicotinamide (15 mM) for 20 min. NAD content was measured using a bioluminescent assay (NAD/NADH-Glo™ assay, [Promega Corporation; Madison, WI, USA])

Determining how Imeglimin's effect on NAD⁺ content is mediated

- The effect of Imeglimin on NAD content was measured in the presence of 16.7 mM glucose for 20 min with and without the addition of gallotannin (10 µM), an inhibitor of nicotinate/nicotinamide mononucleotide adenylyltransferase-1,2,3 (Na/NMNAT-1,2,3) ± Imeglimin (100 µM)
- FK866 (100 nM), an inhibitor of the nicotinamide phosphoribosyltransferase (NAMPT) enzyme, was pre-incubated for 2 h, then incubated for 20 min ± Imeglimin (100 µM). Nicotinamide (15 mM) was used as a positive reference. Phthalic acid (500 µM), an inhibitor of the QPRT enzyme, was pre-incubated for 24 h and then incubated for 20 min ± Imeglimin (100 µM). Quinolinic acid (500 µM) was used as a positive reference

Effect of Imeglimin on insulin release from GK rat islets transfected with CD38 siRNA

- Control siRNA and CD38 siRNA-transfected GK rat islets were incubated for 20 min in the presence of 16.7 mM glucose ± Imeglimin (100 µM). Insulin release was measured by ELISA (Alpco 80-INSRTU-E01)
- Knock-down of CD38 mRNA was confirmed by reverse transcription polymerase chain reaction (RT-PCR) (data not shown)

Effect of Imeglimin on K⁺-ATP channel and phospholipase C (PLC) pathways

- GK rat islets were incubated in 16.7 mM glucose for 20 min, with or without Imeglimin, in the presence or absence of either diazoxide (K⁺-ATP channel opener) to eliminate GSIS via the K⁺-ATP channel, or an inhibitor of PLC (U-73122). Insulin levels were measured by ELISA

Effect of Imeglimin on cyclic adenosine monophosphate (cAMP) content of GK rat islets

- Isolated GK rat islets were pre-incubated for a 30 min stabilization period in the presence of 2.8 mM glucose and then incubated in the presence of 2.8 mM or 16 mM glucose ± Imeglimin (100 µM) for 15 min. cAMP content was measured using the EIA cAMP Biotrack™ assay (GE Healthcare Life Sciences; Little Chalfont, UK)
- GLP-1 (0.1 µM) was used as reference and a phosphodiesterase inhibitor IBMX (1 mM) was added to avoid degradation of cAMP to adenosine monophosphate (AMP)

Results

Imeglimin increased GSIS in islets of diabetic GK rats

- After incubation in 16.7 mM glucose, the presence of Imeglimin (25 to 100 µM) was shown to potentiate insulin secretion in response to glucose in a concentration-dependent manner, with increases at 50 and 100 µM of 35% (p<0.05) and 41% (p<0.01), respectively (Figure 1)

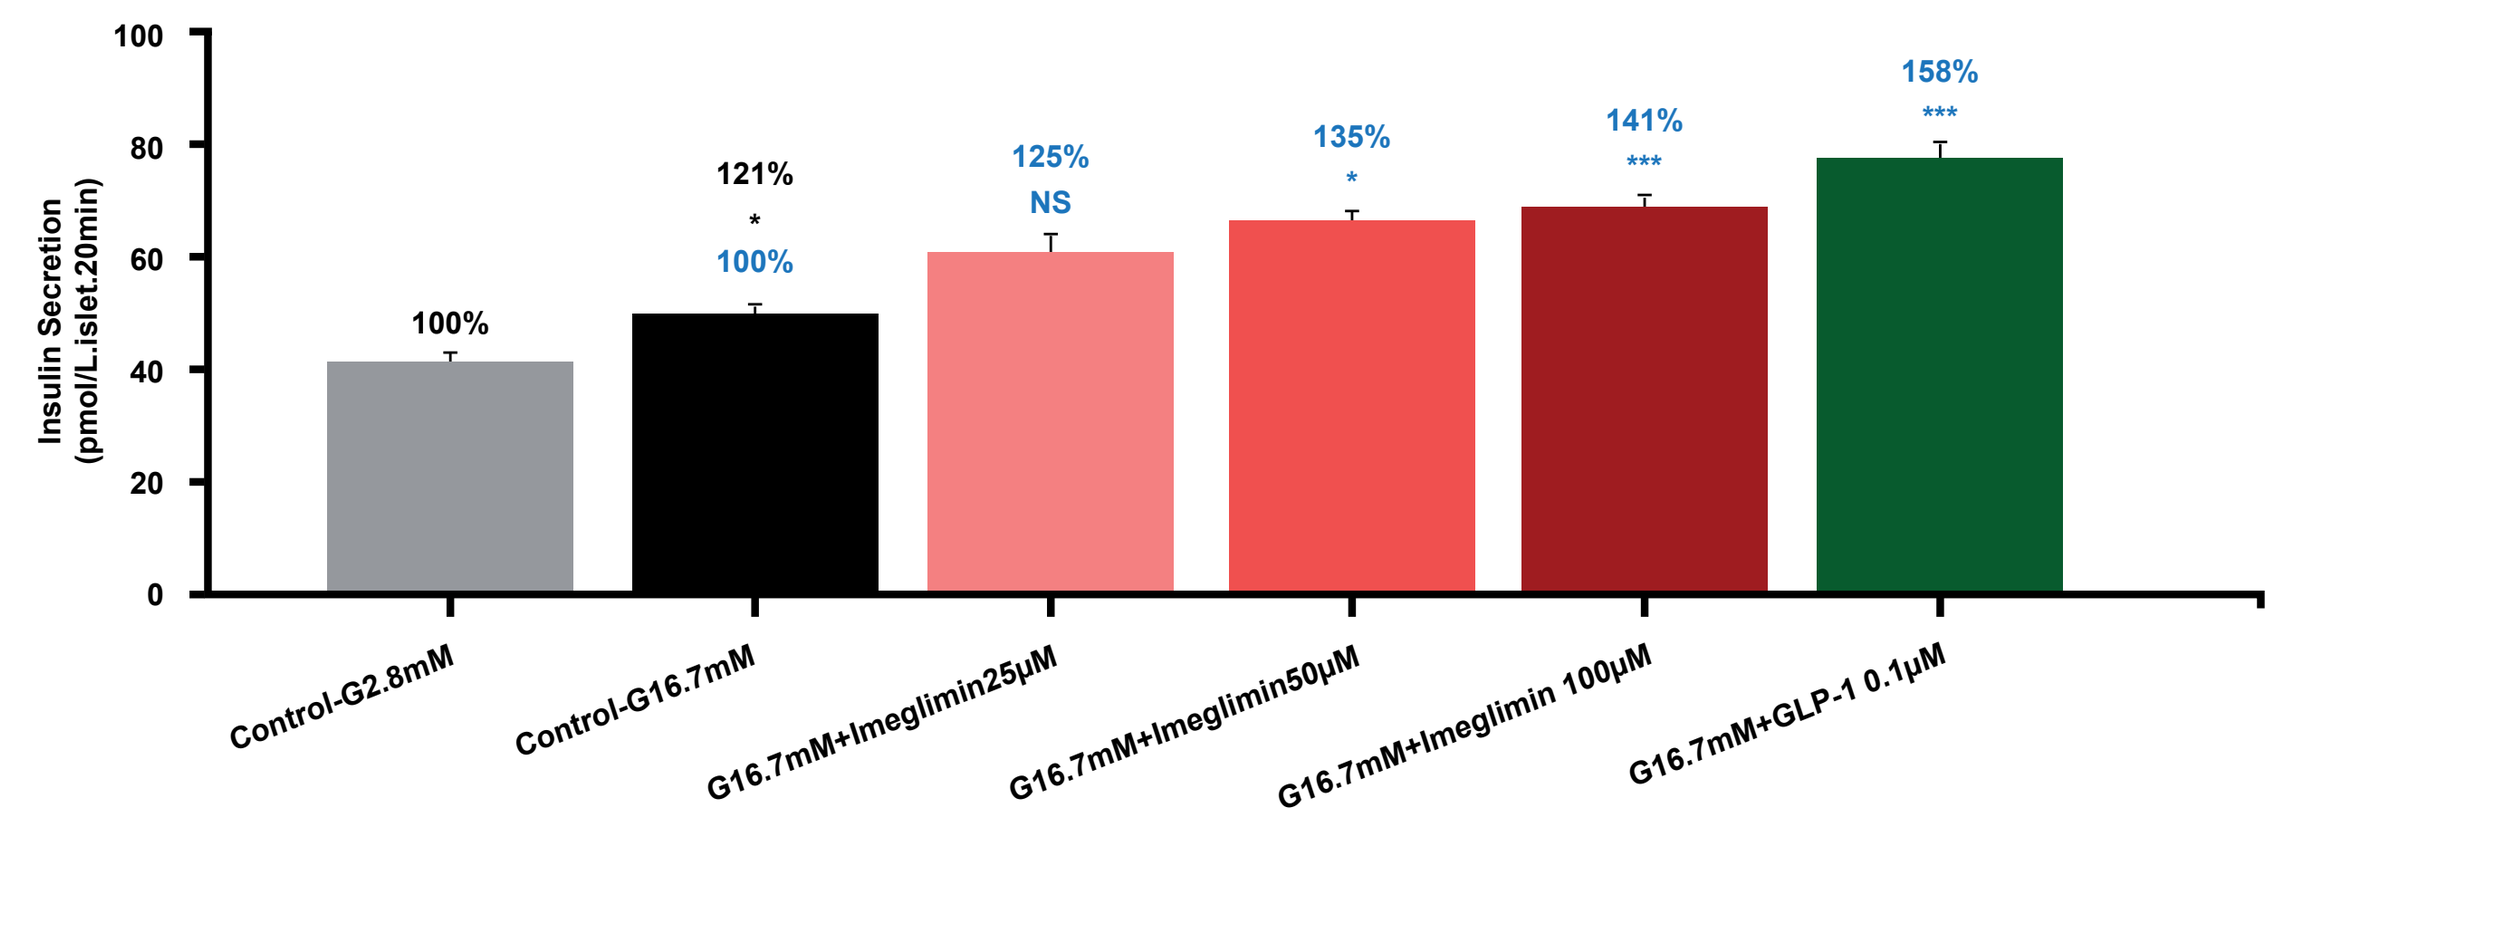
Insulin Secretion (pmol/L.islet.20min)
100%
121% * 100%
125% NS
135% *
141% ***
158% ***
Control-G2.8mM
Control-G16.7mM
G16.7mM+Imeglimin25µM
G16.7mM+Imeglimin50µM
G16.7mM+Imeglimin 100µM
G16.7mM+GLP-1 0.1µM

Figure 1: Imeglimin increased GSIS in islets of diabetic GK rats
*p<0.05, ***p<0.001; GLP-1, glucagon-like peptide-1 agonist; GSIS, glucose-stimulated insulin secretion; NS, not significant

Imeglimin increased NAD content and NAD biosynthesis in islets of diabetic GK rats

- Imeglimin at 100 µM was observed to rapidly induce a 31% increase in NAD (p<0.05) (Figure 2a)
- The addition of gallotannin, an inhibitor of Na/NMNAT-1,2,3 (a key enzyme in both the *de novo* synthesis pathway from tryptophan and salvage pathway from nicotinamide) prevented the increase in NAD content observed with Imeglimin alone (–23% p<0.01), showing that Imeglimin raised NAD content through an increase in NAD synthesis in islets of type 2 diabetic GK rats (Figure 2b)

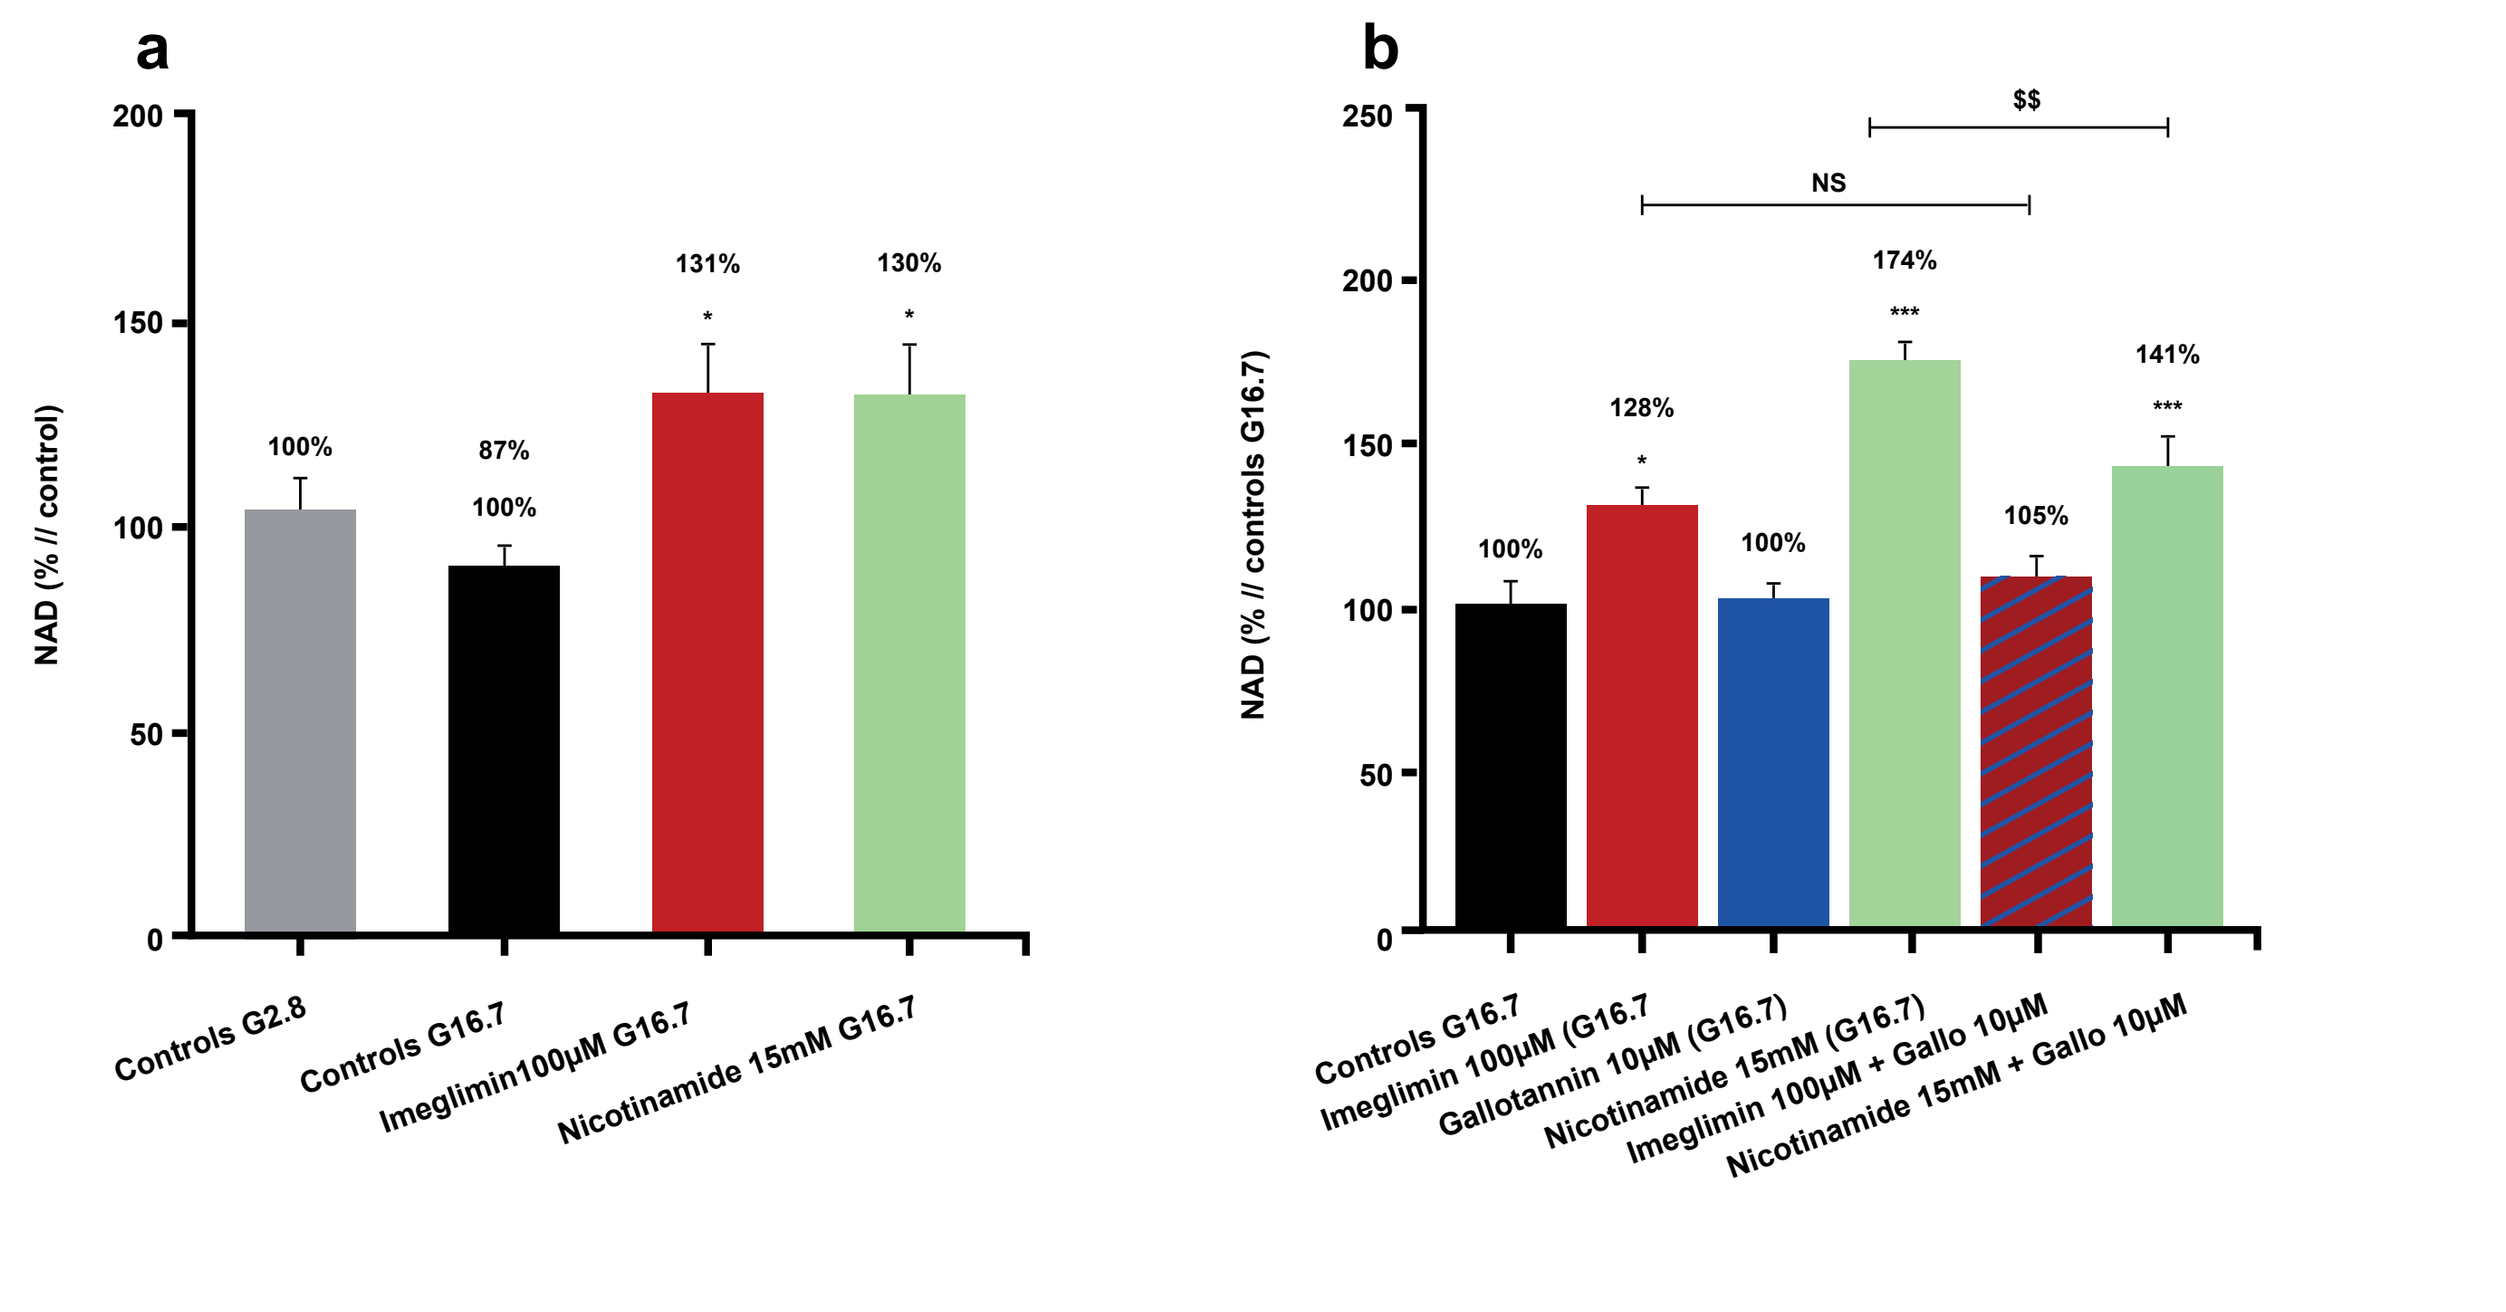
a
NAD (% // control)
100%
87% 100%
131% *
130% *
Controls G2.8
Controls G16.7
Imeglimin100µM G16.7
Nicotinamide 15mM G16.7
b
NAD (% // controls G16.7)
$$
NS
100%
128% *
100%
174% ***
105%
141% ***
Controls G16.7
Imeglimin 100µM (G16.7
Gallotannin 10µM (G16.7)
Nicotinamide 15mM (G16.7)
Imeglimin 100µM + Gallo 10µM
Nicotinamide 15mM + Gallo 10µM

Figure 2a and 2b: Imeglimin increased NAD content and NAD biosynthesis in islets of diabetic GK rats
*p<0.05 vs Imeglimin alone, ***p<0.001 vs Imeglimin alone; $$ p<0.01 vs nicotinamide alone; NAD, nicotinamide adenine dinucleotide; NS, not significant

The increase in NAD⁺ content caused by Imeglimin is dependent on the salvage pathway

- FK866, an inhibitor of the NAMPT enzyme, is key in the salvage pathway for NAD synthesis. Islets incubated with phthalic acid (inhibitor of the QPRT enzyme) alone, which is key for the *de novo* synthesis of NAD, had no effect on NAD content in GK rat islets, whereas the NAD precursor nicotinamide (reference of salvage pathway) increased NAD content in rat islets after 20 min incubation
- In GK rat islets, the co-incubation of Imeglimin and phthalic acid, which inhibits the QPRT enzyme in the *de novo* pathway, induced a similar increase in NAD content to Imeglimin alone even if the difference did not reach significance compared with control value (33% and 25%, respectively; not significant), showing no involvement of *de novo* pathway in Imeglimin effect
- In GK rat islets in the presence of Imeglimin and FK866, which inhibits the NAMPT enzyme in the salvage pathway, a significant 23% decrease (p<0.01) was observed compared with Imeglimin alone
- The lack of increase in NAD content when the NAMPT enzyme is inhibited suggests that the salvage pathway is involved in the effect of Imeglimin on NAD increase (Figure 3a and 3b)

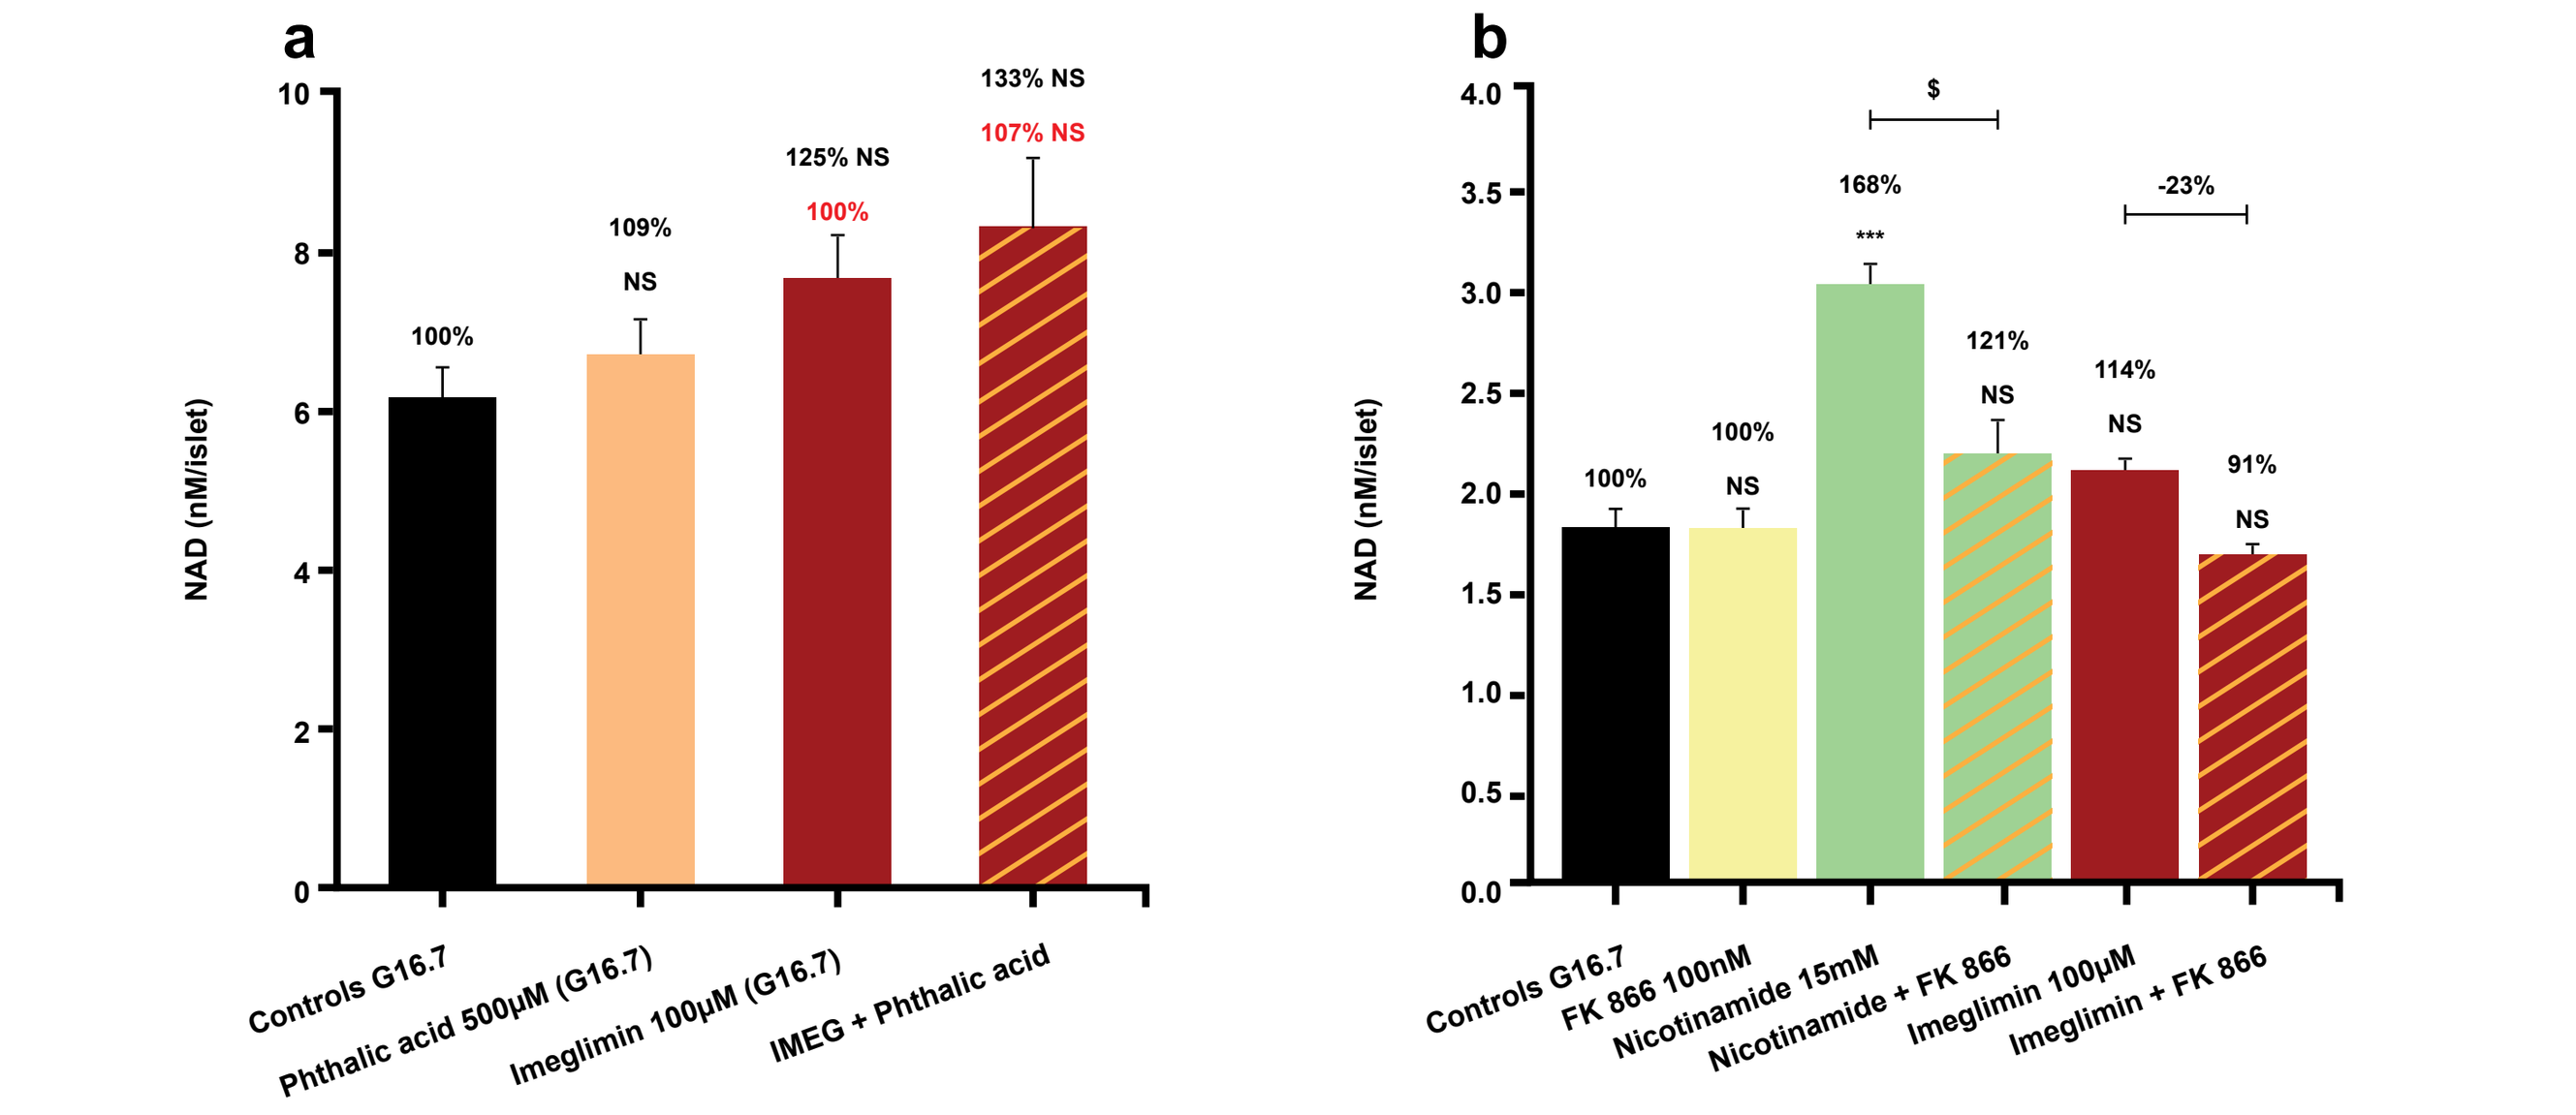
a
NAD (nM/islet)
100%
109% NS
125% NS 100%
133% NS 107% NS
Controls G16.7
Phthalic acid 500µM (G16.7)
Imeglimin 100µM (G16.7)
IMEG + Phthalic acid
b
NAD (nM/islet)
$
-23%
100%
100% NS
168% ***
121% NS
114% NS
91% NS
Controls G16.7
FK 866 100nM
Nicotinamide 15mM
Nicotinamide + FK 866
Imeglimin 100µM
Imeglimin + FK 866

Figure 3a and 3b: Increase in NAD+ content caused by Imeglimin is dependent on the salvage pathway
***p<0.001 vs G16.7 controls; $ p<0.05 vs nicotinamide alone; -23% p<0.01 vs Imeglimin alone; IMEG, Imeglimin; NAD, nicotinamide adenine dinucleotide; NS, not significant

CD38 involvement is mandatory to the effect of Imeglimin on GSIS

- CD38 catalyzes the conversion of NAD into active metabolites involved in Ca^{2+} mobilization that leads to insulin secretion
- In islets treated with CD38 siRNA, Imeglimin had no significant effect on GSIS, whereas in islets treated with control siRNA, Imeglimin treatment was observed to lead to a 51% increase in insulin release (p<0.05), suggesting that NAD metabolites have an important role in Imeglimin's MOA (Figure 4)

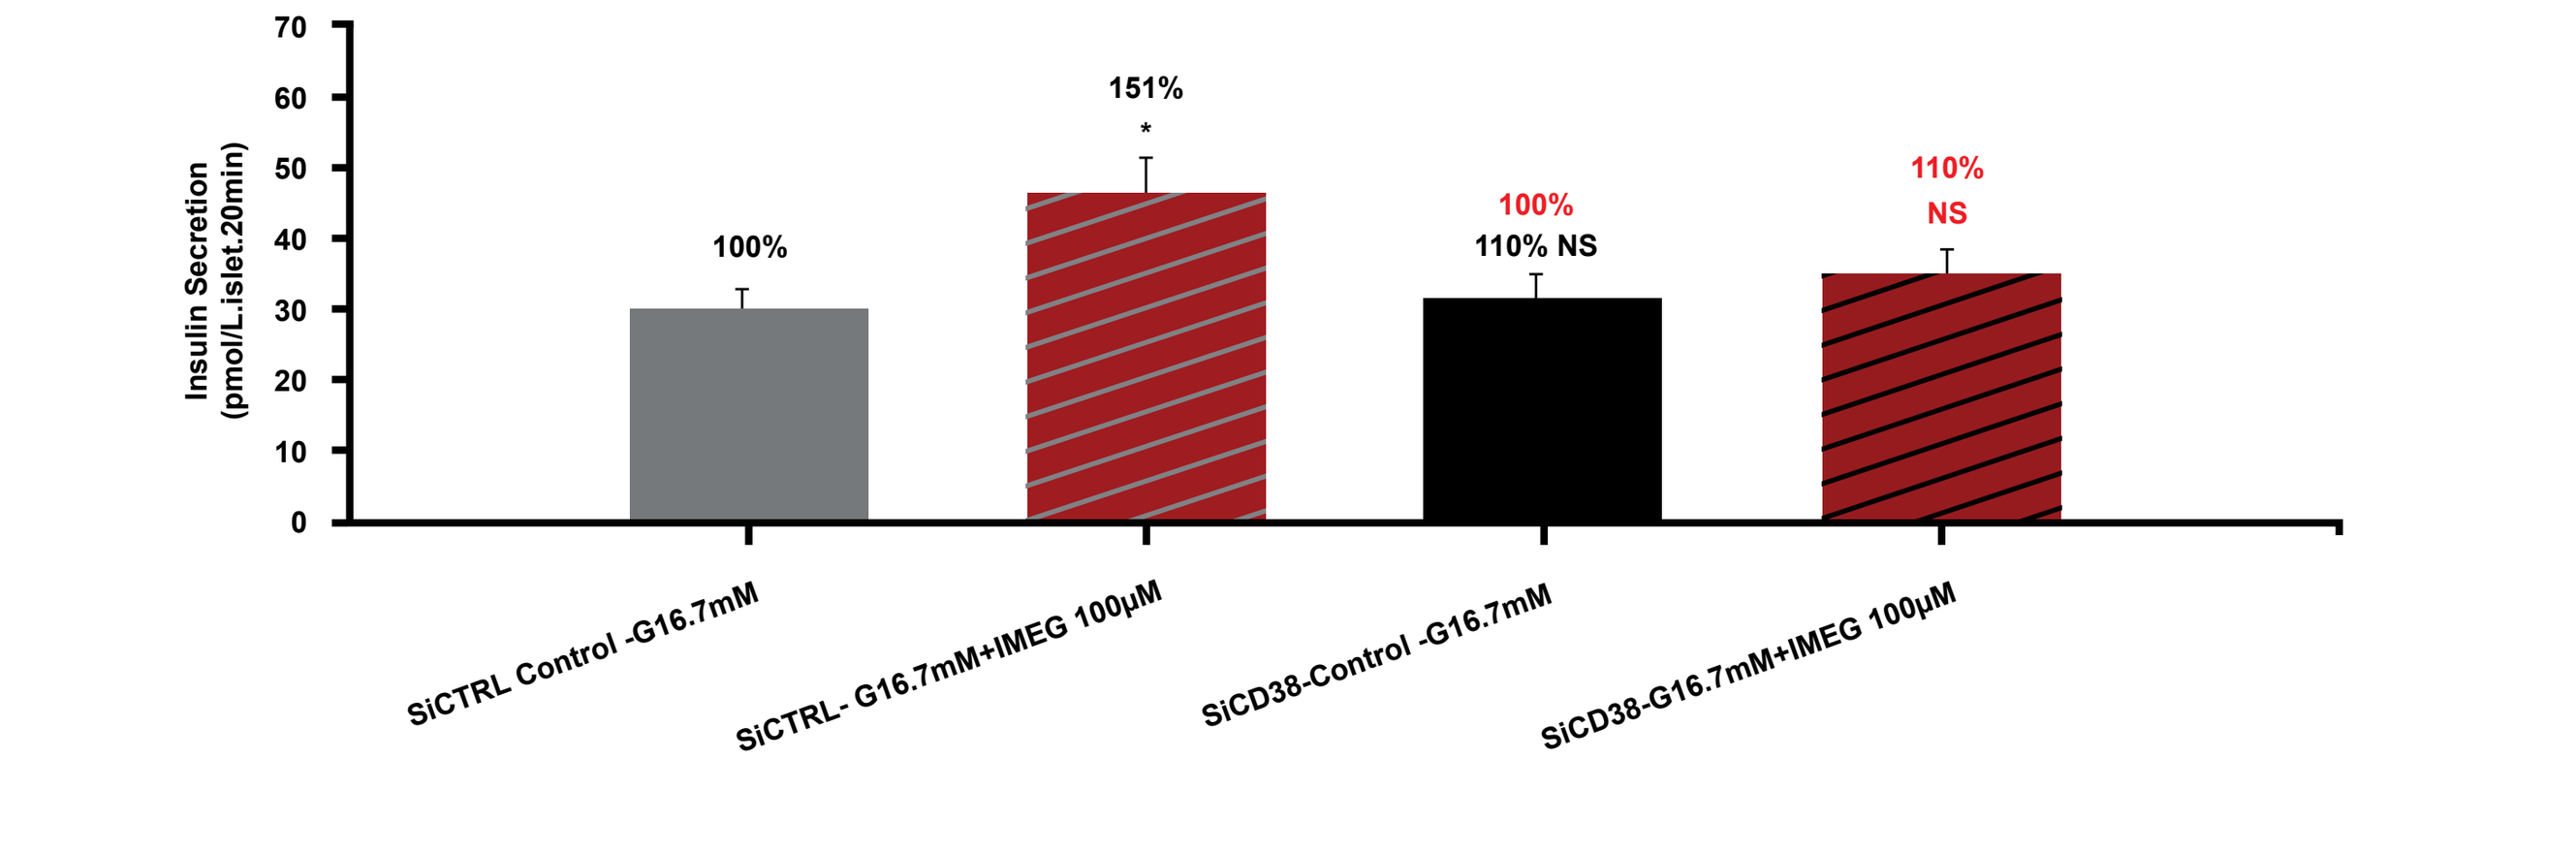
Insulin Secretion (pmol/L.islet.20min)
100%
151% *
100% 110% NS
110% NS
SiCTRL Control -G16.7mM
SiCTRL- G16.7mM+IMEG 100µM
SiCD38-Control -G16.7mM
SiCD38-G16.7mM+IMEG 100µM

Figure 4: CD38 involvement is mandatory to the effect of Imeglimin on GSIS
*p<0.05; IMEG, Imeglimin; NS, not significant; siCD38, CD38 siRNA; siCTRL, control siRNA

K⁺-ATP channels and PLC pathway were not involved in Imeglimin's potentiating action on GSIS

- The co-incubation of the K⁺-ATP channel opener diazoxide with Imeglimin did not modify the potentiating effect on glucose-induced insulin secretion observed with Imeglimin alone
- The inhibition of the PLC pathway with U-73122 did not significantly alter the effect of Imeglimin on insulin secretion compared with Imeglimin alone in 16.7 mM glucose
- Both results suggest that neither K⁺-ATP channels nor the PLC pathway are involved in Imeglimin's potentiating effect on GSIS (Figure 5a and 5b)

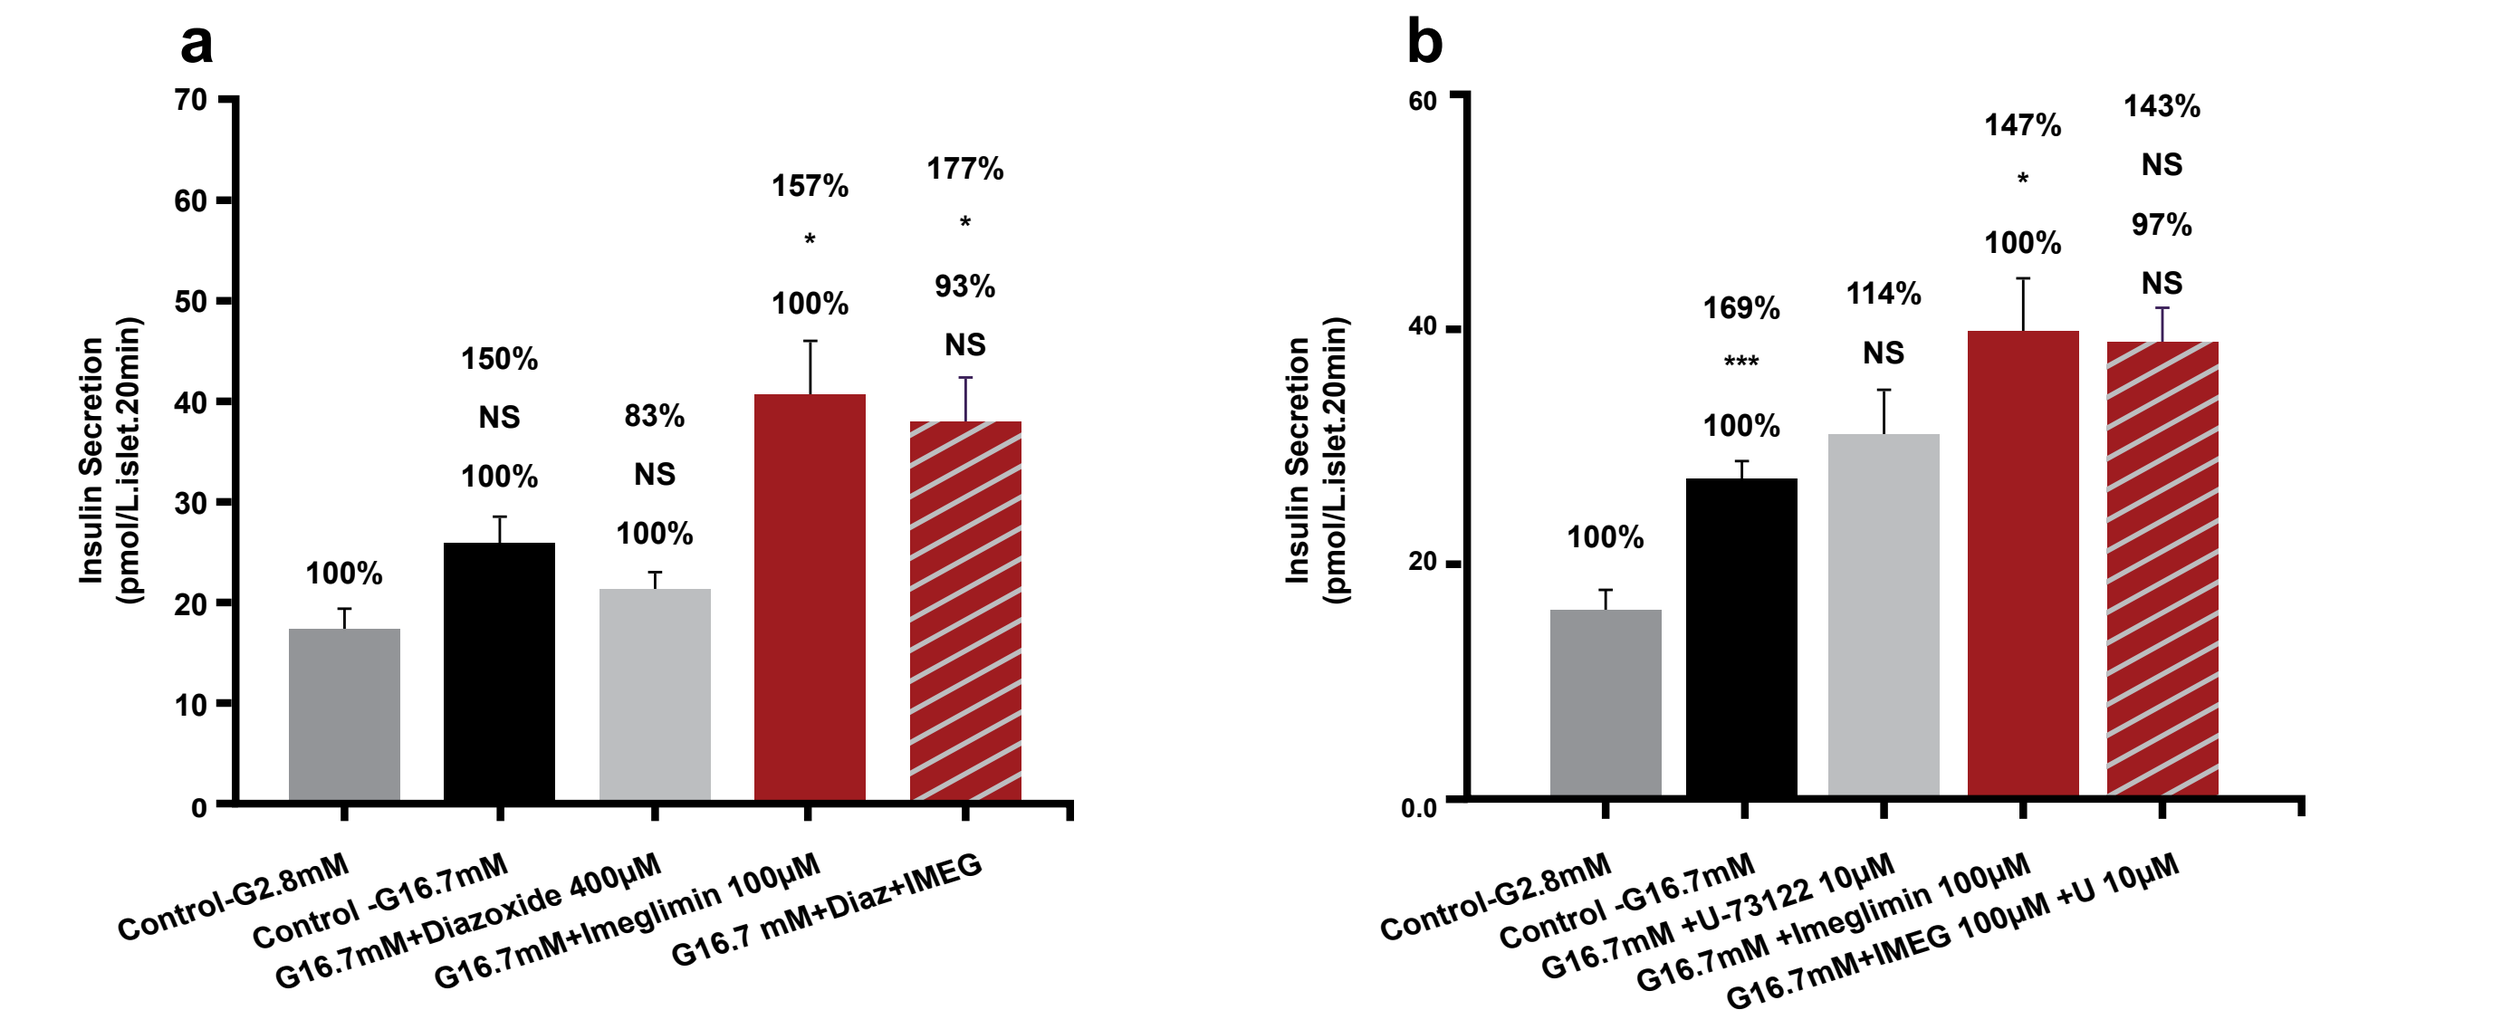
a
Insulin Secretion (pmol/L.islet.20min)
100%
150% NS 100%
83% NS 100%
157% * 100%
177% * 93% NS
Control-G2.8mM
Control -G16.7mM
G16.7mM+Diazoxide 400µM
G16.7mM+Imeglimin 100µM
G16.7 mM+Diaz+IMEG
b
Insulin Secretion (pmol/L.islet.20min)
100%
169% *** 100%
114% NS
147% * 100%
143% NS 97% NS
Control-G2.8mM
Control -G16.7mM
G16.7mM +U-73122 10µM
G16.7mM +Imeglimin 100µM
G16.7mM+IMEG 100µM +U 10µM

Figure 5a and 5b: K+-ATP channels and PLC pathway were not involved in Imeglimin's potentiating action on GSIS
*p<0.05; ***p<0.001 vs respective control value;
Diaz, diazoxide; GSIS, glucose-stimulated insulin secretion; IMEG, Imeglimin; NS, not significant

cAMP pathway was not involved in Imeglimin's potentiating action on GSIS

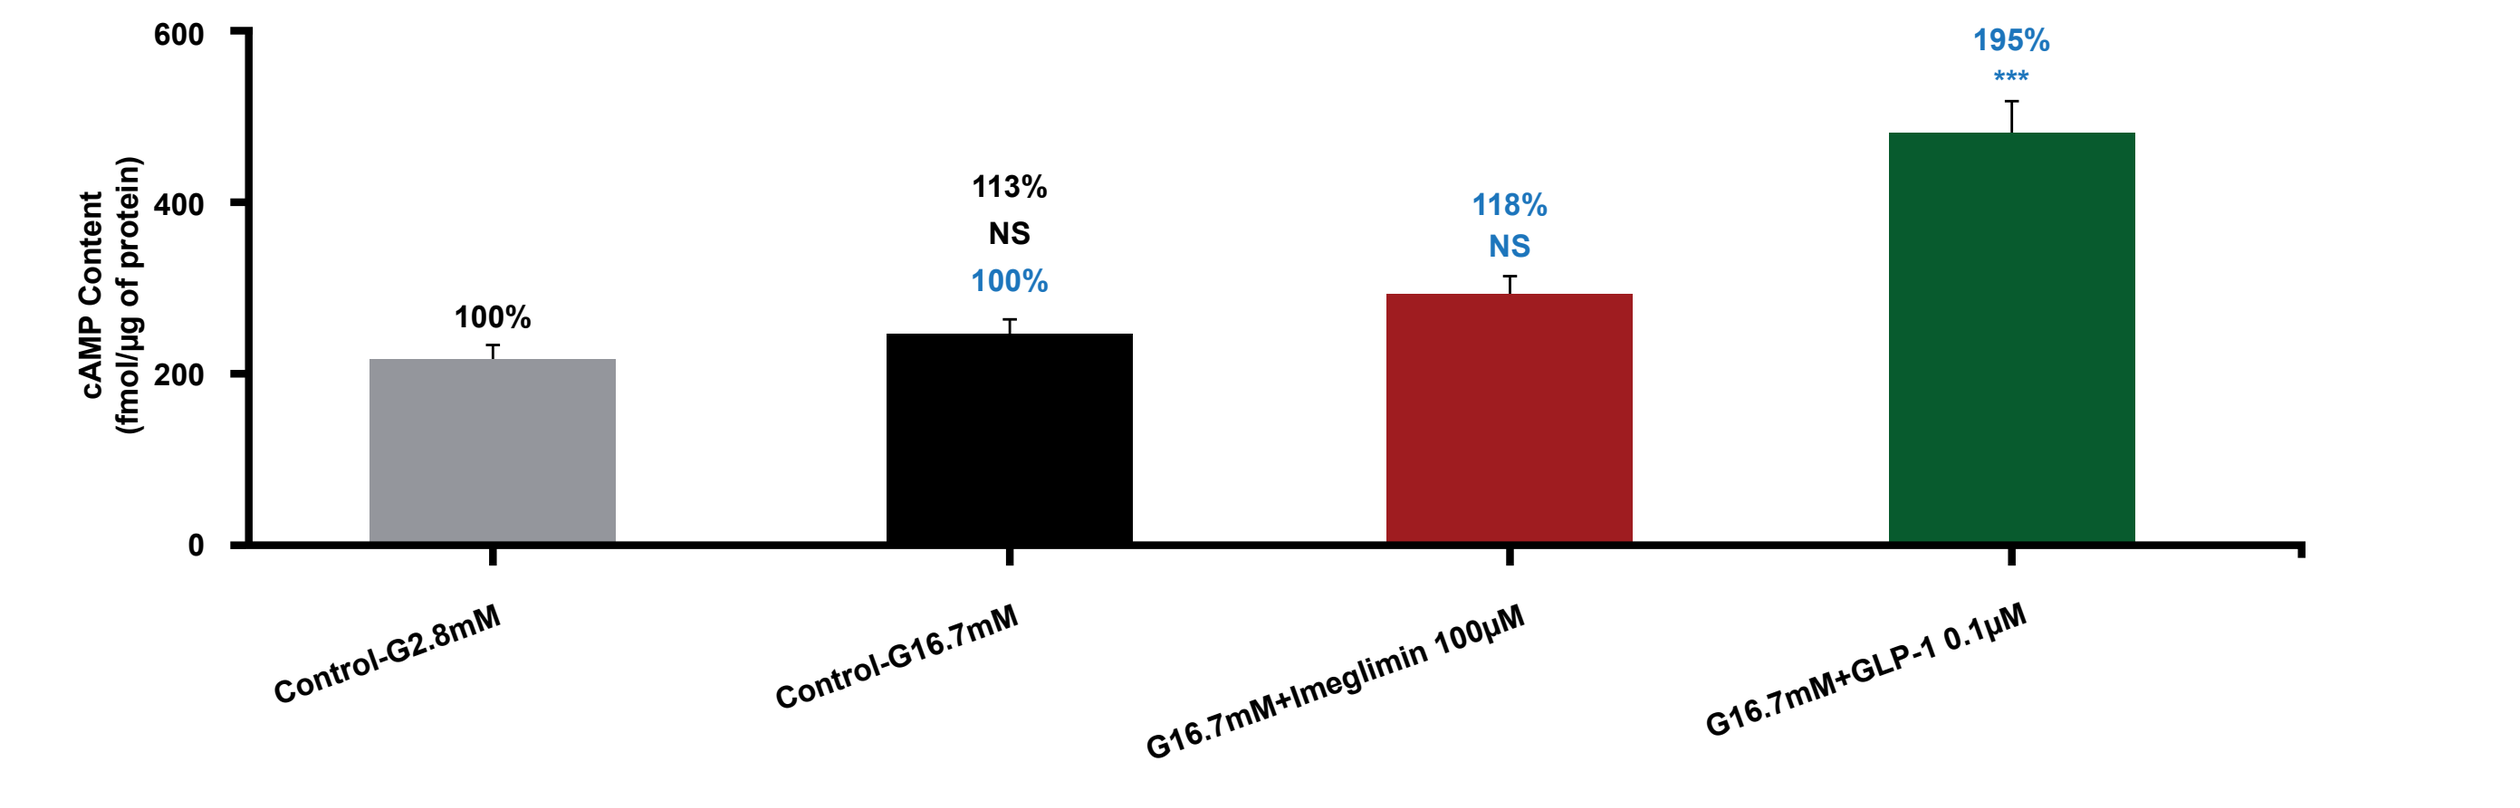
cAMP Content (fmol/µg of protein)
100%
113% NS 100%
118% NS
195% ***
Control-G2.8mM
Control-G16.7mM
G16.7mM+Imeglimin 100µM
G16.7mM+GLP-1 0.1µM

Figure 6: cAMP pathway was not involved in Imeglimin's potentiating action on GSIS
***p<0.001 vs respective control value
cAMP, cyclic adenosine monophosphate; GLP-1, glucagon-like peptide-1 agonist; GSIS, glucose-stimulated insulin secretion; NS, not significant

- In the presence of Imeglimin (100 µM), no significant increase in cAMP content was observed after 15 min incubation (Figure 6)

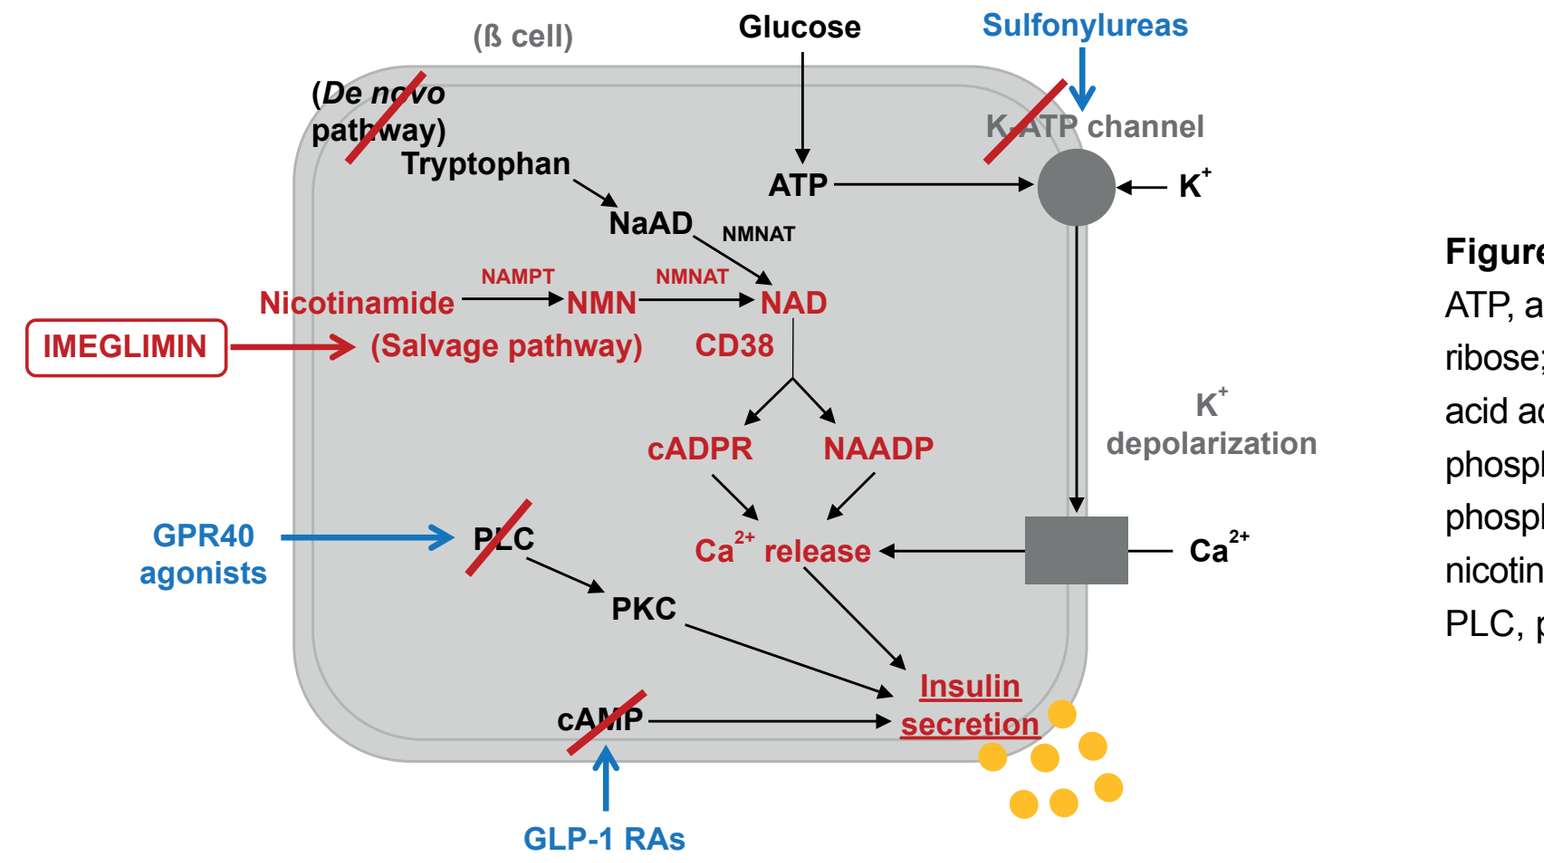
(ß cell)
Glucose
Sulfonylureas
(De novo pathway)
Tryptophan
K-ATP channel
ATP
K⁺
NaAD
NMNAT
NAMPT
NMNAT
Nicotinamide
NMN
NAD
IMEGLIMIN
(Salvage pathway)
CD38
K⁺ depolarization
cADPR
NAADP
GPR40 agonists
PLC
Ca²⁺ release
Ca²⁺
PKC
Insulin secretion
cAMP
GLP-1 RAs

Figure 7: Proposed insulin secretion mechanism of Imeglimin
ATP, adenosine triphosphate; cADPR, cyclic adenosine disphosphate ribose; GLP-1 RA, glucagon-like peptide 1 receptor agonist; NaAD, nicotinic acid adenine dinucleotide; NAADP, nicotinic acid adenine dinucleotide phosphate; NAD, nicotinamide adenine dinucleotide; NAMPT, nicotinamide phosphoribosyltransferase; NMN, β-nicotinamide mononucleotide; NMNAT, nicotinamide mononucleotide adenylyltransferase; PKC, protein kinase C; PLC, phospholipase C

Conclusion

- Imeglimin was observed to significantly and dose-dependently increase GSIS in isolated islets of diabetic GK rats
- This effect is driven by an increase in NAD content
- This increase in NAD content arises due to an increase in NAD synthesis that comes from nicotinamide through the salvage pathway, but not from the *de novo* tryptophan pathway
- Imeglimin's potentiating action on GSIS does not involve K⁺-ATP channels, PLC and cAMP as confirmed with the use of specific inhibitors of the main GSIS pathways (Figure 7)
- Taken together, these results demonstrate that Imeglimin's MOA leads to a potentiation of GSIS in diabetic islets that is novel and different from known insulin secretagogue agents

Conflicts of Interest

Sophie Hallakou-Bozec and Sebastien Bolze are Poxel employees.

Micheline Kergoat is an employee of Metabrain Research. Metabrain Research performed these experiments.

References

1. Pirags V, et al. *Diabetes Obes Metab* 2012;14:852–858; 2. Vial G, et al. *Diabetes* 2015;64:2254–2264; 3. Pacini G, et al. *Diabetes Obes Metab* 2015;17:541–545

Corresponding author: Sophie Bozec, 259-261 avenue Jean Jaurès, 69007 Lyon, France
Email: sophie.bozec@poxelpharma.com