1197-P American Diabetes Association, 10th–14th June, New Orleans, LA, USA

Imeglimin Improves Insulin Sensitivity in an Adult STZ Rat Model

Sophie Hallakou-Bozec[1], Sebastien Bolze[1], Micheline Kergoat[2], Michael Roden[3–5], Harold E. Lebovitz[6]

[1]Poxel SA, 259-261 avenue Jean Jaurès, 69007 Lyon, France; [2]Metabrain Research, 4 avenue du Président François Mitterrand, 91380 Chilly–Mazarin, France; [3]Institute for Clinical Diabetology, German Diabetes Center, Leibniz Institute for Diabetes Research, Auf'm Hennekamp 65, 40225, Düsseldorf, Germany; [4]German Center for Diabetes Research (DZD e.V.), Partner Düsseldorf, Germany; [5]Department of Endocrinology and Diabetology, Medical Faculty, Heinrich Heine University Düsseldorf, Düsseldorf, Germany; [6]State University of New York, Health Sciences Center, Brooklyn, NY, USA

Abstract

Imeglimin is the first in a new tetrahydrotriazine-containing class of oral glucose-lowering agents, the glimins. It targets two critical metabolic defects at the root of type 2 diabetes: impaired insulin secretion and insulin resistance. Imeglimin's beneficial effect on insulin sensitivity during an insulin tolerance test was shown in a high-fat, high-sucrose diet-fed mouse model and further through improvement in sensitivity indexes during oral glucose tolerance tests in patients with type 2 diabetes. This study describes the effect of Imeglimin on insulin sensitivity during a euglycemic hyperinsulinemic clamp in streptozotocin (STZ)-injected rats.

Ten days post-STZ injection (60 mg/kg intraperitoneal), 12-week-old diabetic rats were treated with Imeglimin (150 mg/kg bid PO) for 15 days. After both acute and 10 days of Imeglimin administration, oral glucose tolerance was significantly improved in overnight-fasted STZ rats ($AUC_{(T0)}$ glucose: –41% and –35%, respectively; p<0.001).

Hepatic glucose production rate (GPR) and whole-body glucose utilization rates (GUR) were evaluated both in the basal state and during euglycemic hyperinsulinemic clamp (insulin: 0.5 U/h/kg) performed in overnight-fasted STZ rats after 15 days of Imeglimin treatment. No significant changes of GPR and GUR were observed in the basal state. During the clamp, at similar levels of euglycemia and hyperinsulinemia, the steady-state glucose infusion rate (SSGIR) increased by 209% in STZ rats after Imeglimin treatment as compared with the SSGIR of the control rats (p<0.01). Imeglimin treatment was also observed to significantly decrease hepatic GPR (–40%; p<0.05). GUR was increased, although not significantly, in the Imeglimin-treated group (29%).

Imeglimin induced a potent beneficial effect on glucose tolerance after both acute and chronic treatment (10 days) in overnight-fasted adult STZ rats. In this model of deep insulinopenia, we demonstrated that Imeglimin clearly improved hepatic insulin sensitivity during the euglycemic hyperinsulinemic clamp. We have previously demonstrated that Imeglimin increases glucose-stimulated insulin secretion (GSIS) in various models, as well as in patients with type 2 diabetes. This study supports previous observations of the effects of Imeglimin on insulin sensitivity and demonstrates its unique mechanism of action (MOA) targeting both insulin secretion and insulin sensitivity.

Background

- Imeglimin is the first in a new tetrahydrotriazine-containing class of oral glucose-lowering agents, the glimins[1]
- The unique MOA of Imeglimin involves the regulation of mitochondrial bioenergetics as observed in a high-fat, high-sucrose diet-fed mouse model[2]
- Imeglimin has been observed to decrease hepatic GPR and β-cell apoptosis and to increase skeletal muscle glucose uptake[3]
- Imeglimin's MOA leads to a potentiation of GSIS, as previously demonstrated *in vivo* in animal models of diabetes and in patients with type 2 diabetes[4]

Objectives

- The aim of this study was to evaluate the effects of Imeglimin on glucose tolerance and hepatic and peripheral insulin sensitivity in an adult STZ rat model.

Research Methods

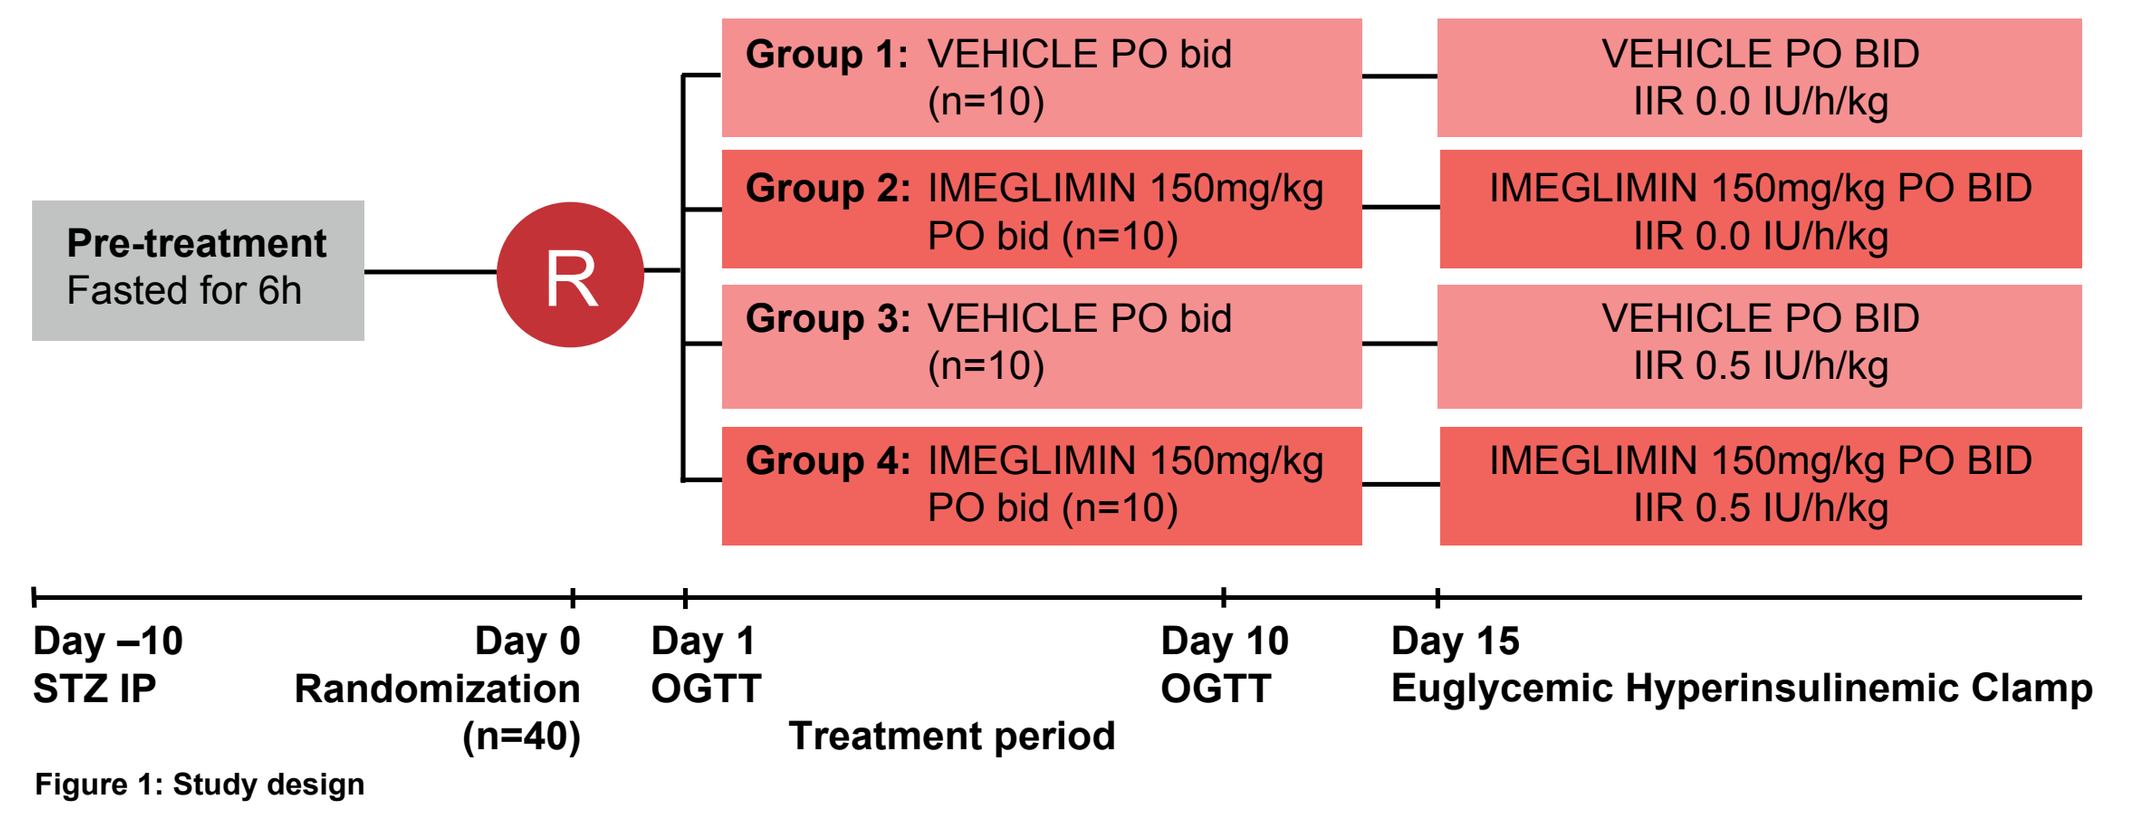
Pre-treatment
Fasted for 6h
R
Group 1: VEHICLE PO bid (n=10)
Group 2: IMEGLIMIN 150mg/kg PO bid (n=10)
Group 3: VEHICLE PO bid (n=10)
Group 4: IMEGLIMIN 150mg/kg PO bid (n=10)
VEHICLE PO BID IIR 0.0 IU/h/kg
IMEGLIMIN 150mg/kg PO BID IIR 0.0 IU/h/kg
VEHICLE PO BID IIR 0.5 IU/h/kg
IMEGLIMIN 150mg/kg PO BID IIR 0.5 IU/h/kg
Day –10 STZ IP
Day 0 Randomization (n=40)
Day 1 OGTT
Treatment period
Day 10 OGTT
Day 15 Euglycemic Hyperinsulinemic Clamp

Figure 1: Study design

BID, twice daily; IIR, insulin infusion rate; IP, intraperitoneal; IU, international units; OGTT, oral glucose tolerance test; PO, per os; R, randomization; STZ, streptozotocin

- Six-hour-fasted rats were given an injection of STZ (60mg/kg IP), resulting in a high level of 3-hour-fasted glycemia (24mM)
- After one week, the rats were randomized into 4 groups (n=10/group) on the basis of their glucose level after 3 hours of fasting (Figure 1)
- Ten days after the STZ injection, rats were treated twice daily for 2 weeks with Imeglimin or vehicle
- On the first day of treatment and on Day 10 of the study, administration of Imeglimin or vehicle was carried out in all groups of overnight-fasted rats. The first blood sample was collected 1 hour after treatment at T0 just before the oral glucose load, and other samples were collected at T+10, T+20, T+30, T+60, and T+120 min after the glucose load. All plasma samples were frozen and stored at –20°C prior to glucose and insulin measurement
- On Day 15, the last day of treatment, a euglycemic hyperinsulinemic clamp was performed 45 min after the last administration of Imeglimin or vehicle in diabetes-induced overnight-fasted rats in order to study both endogenous GPR and total glucose utilization
- In this study, basal state insulin (groups 1 and 2) and moderate hyperinsulinemia induced by one insulin infusion (groups 3 and 4) were tested. During the clamp, plasma glucose was maintained at a constant euglycemic level by varying the infusion of exogenous glucose (12.5%). Endogenous GPR and glucose turnover in the basal state and during clamp studies were assessed with a primed continuous infusion of [3-3H]-glucose
- The rate of glucose disposal (i.e. GUR) was calculated as GUR = Rd (rate of glucose disappearance), and the hepatic GPR as GPR = Rate of glucose appearance (Ra) – SSGIR

Results

Imeglimin observed to decrease basal glycemia and improve glucose tolerance after both acute and chronic administration

At Day 1:

- Acute oral treatment of Imeglimin was observed to decrease fasting glycemia by 23% as compared with control rats (p<0.001); area under the curve of the glycemic response to a glucose challenge decreased by 41% in Imeglimin-treated rats (p<0.001) as compared with control rats (Figure 2a and 2b)

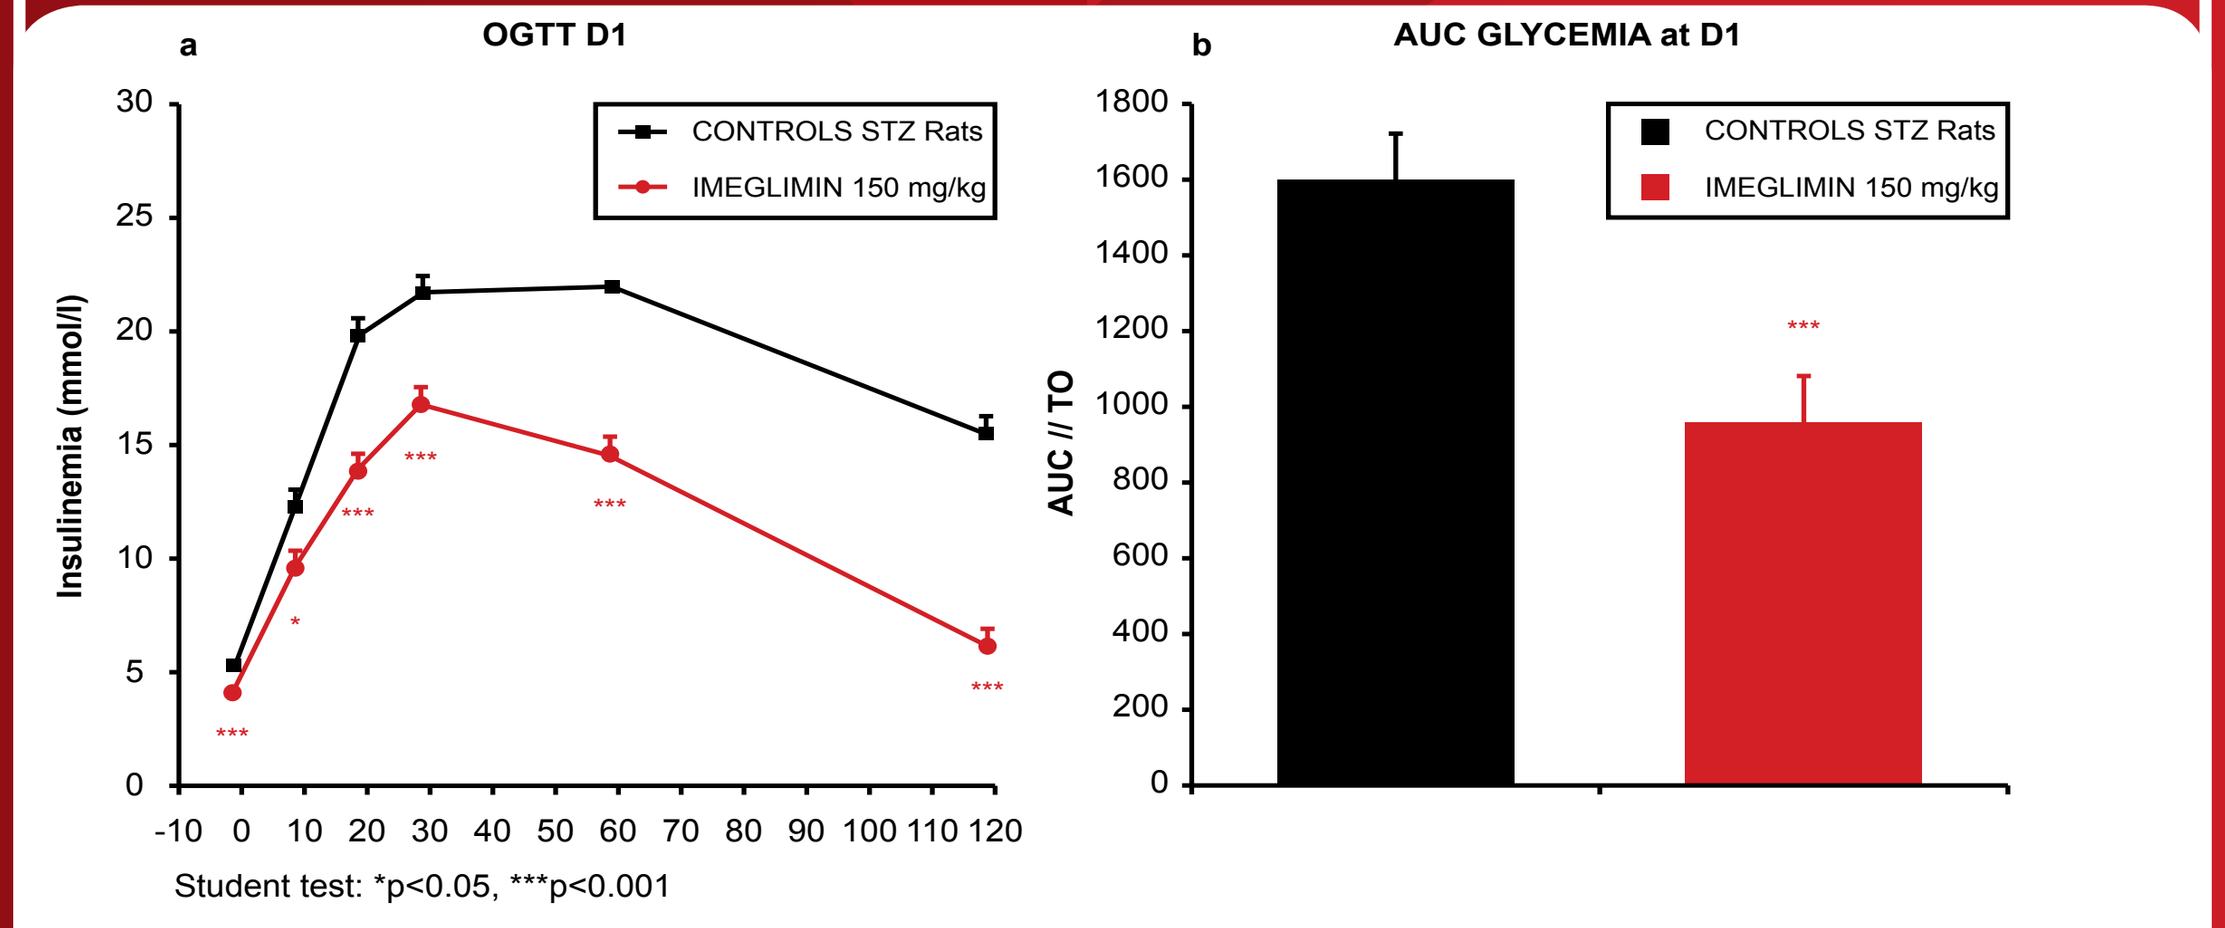
a
OGTT D1
CONTROLS STZ Rats
IMEGLIMIN 150 mg/kg
Insulinemia (mmol/l)
Student test: *p<0.05, ***p<0.001
b
AUC GLYCEMIA at D1
AUC // TO

Figure 2a and 2b
AUC, area under curve; D1, treatment day 1; OGTT, oral glucose tolerance test; STZ, streptozotocin

- A single dose of Imeglimin did not modify fasting insulinemia of the STZ rats. However, a 76% increase in insulin secretion in response to glucose challenge (p<0.001) was observed as compared with the control rats (Figure 3a and 3b)

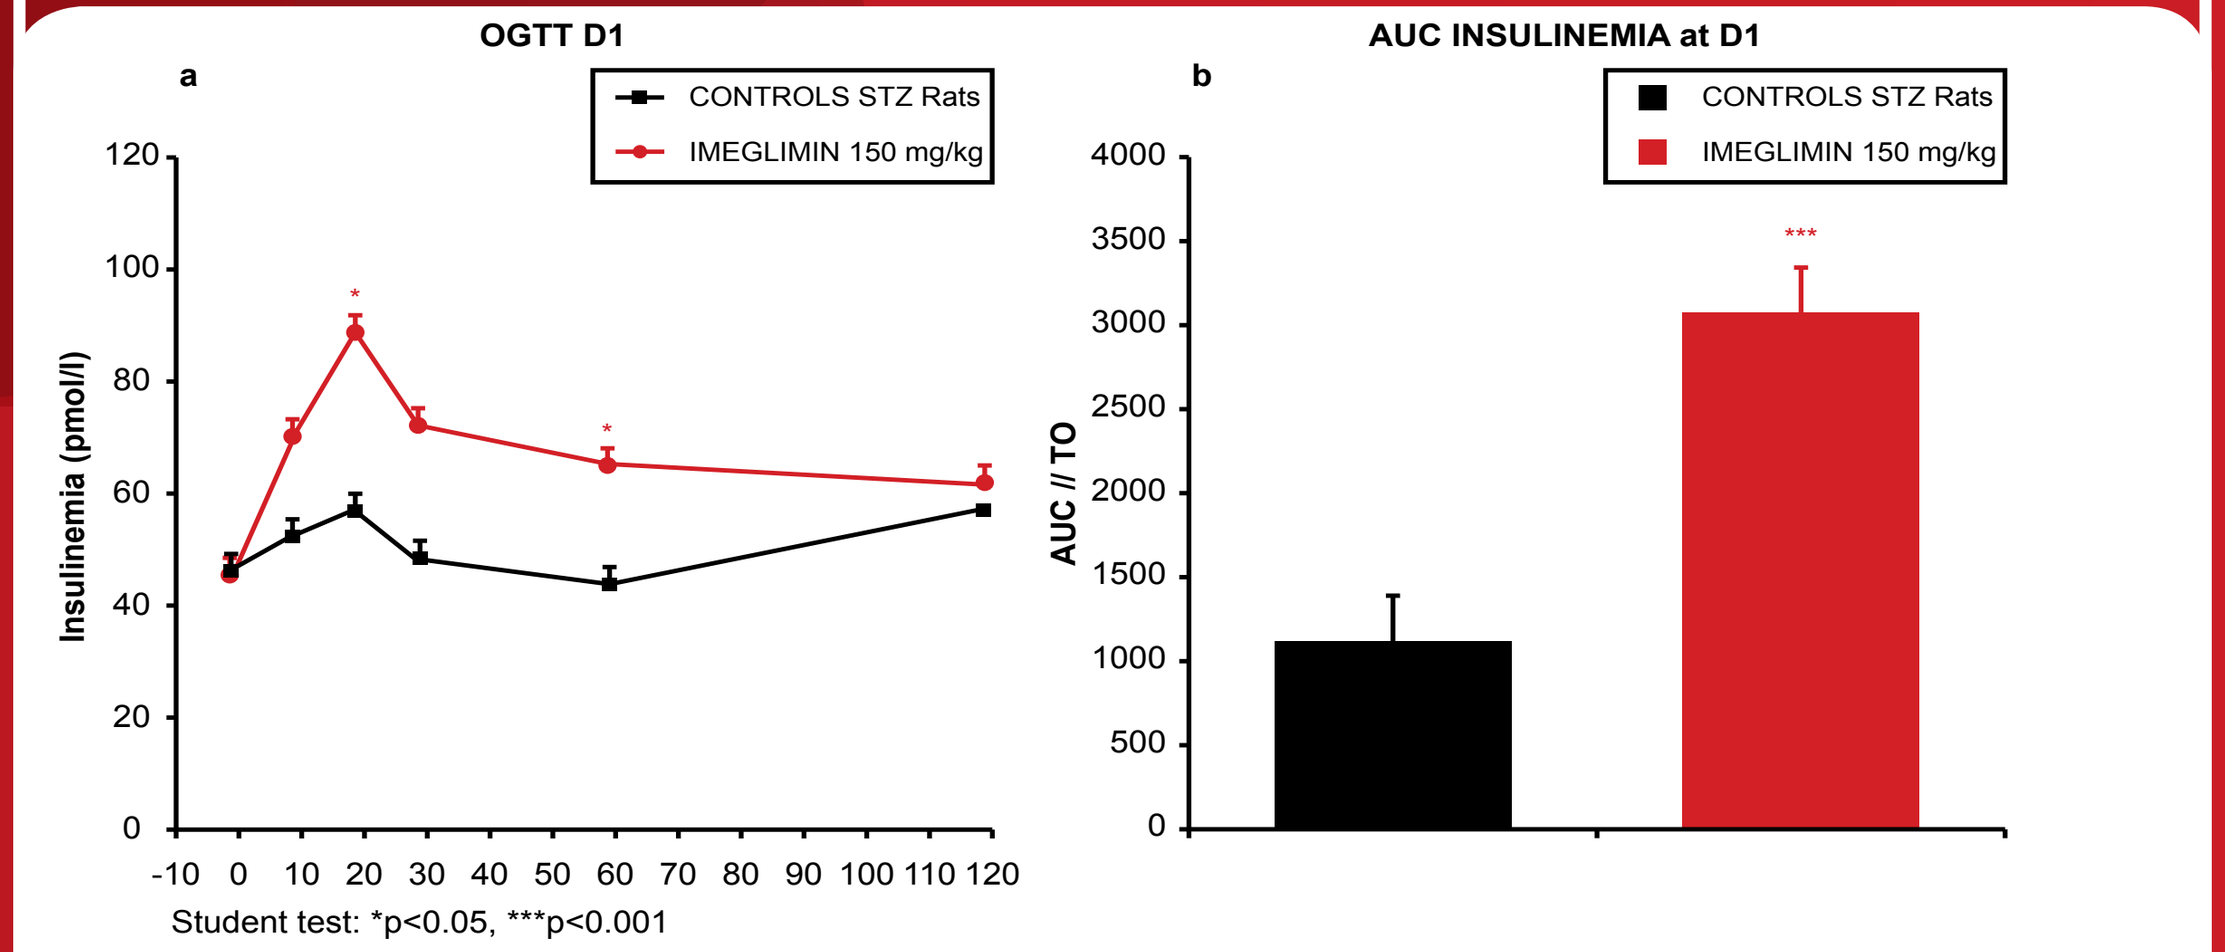
a
OGTT D1
CONTROLS STZ Rats
IMEGLIMIN 150 mg/kg
Insulinemia (pmol/l)
Student test: *p<0.05, ***p<0.001
b
AUC INSULINEMIA at D1
AUC // TO

Figure 3a and 3b
AUC, area under curve; D1, treatment day 1; OGTT, oral glucose tolerance test; STZ,streptozotocin

At Day 10:

- Oral treatment of Imeglimin twice a day for 10 days significantly reduced fasting glycemia (–29%; p<0.05) and improved glucose tolerance
- Area under the curve of the glycemic response to a glucose challenge was 35% lower in Imeglimin-treated rats compared with control rats (p<0.001) (Figure 4a and 4b)

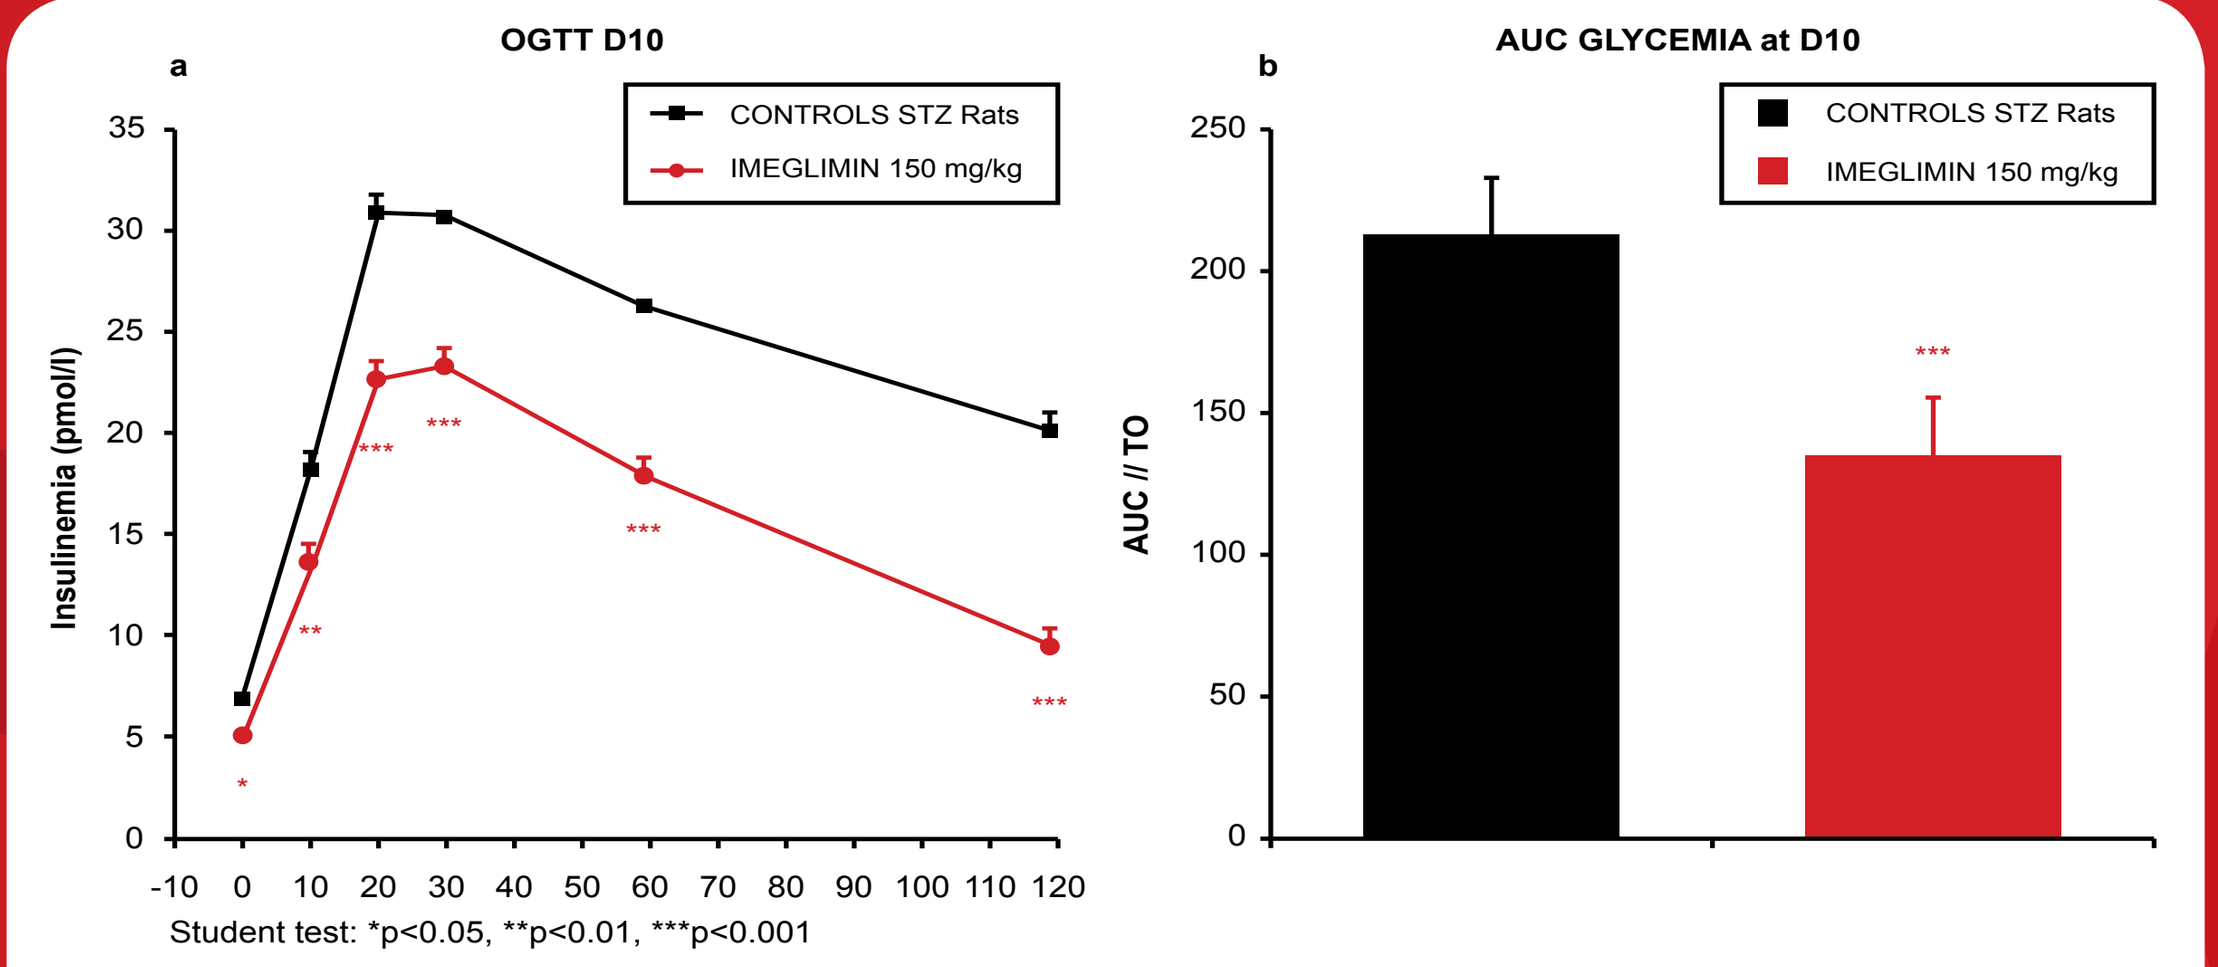
a
OGTT D10
CONTROLS STZ Rats
IMEGLIMIN 150 mg/kg
Insulinemia (pmol/l)
Student test: *p<0.05, **p<0.01, ***p<0.001
b
AUC GLYCEMIA at D10
AUC // TO

Figure 4a and 4b
AUC, area under curve; D10, treatment day 10; OGTT, oral glucose tolerance test; STZ,streptozotocin

- Fasting insulinemia in control STZ rats decreased by 48% at Day 10 as compared with Day 1 as a result of β-cell destruction induced by STZ injection. Imeglimin significantly increased basal insulinemia (+42%; p<0.05) and insulin secretion in response to glucose challenge ($AUC_{(T0)}$: +216%; p<0.05), suggesting a preservation of β-cell function and/or mass in Imeglimin-treated STZ rats (Figure 5a and 5b)

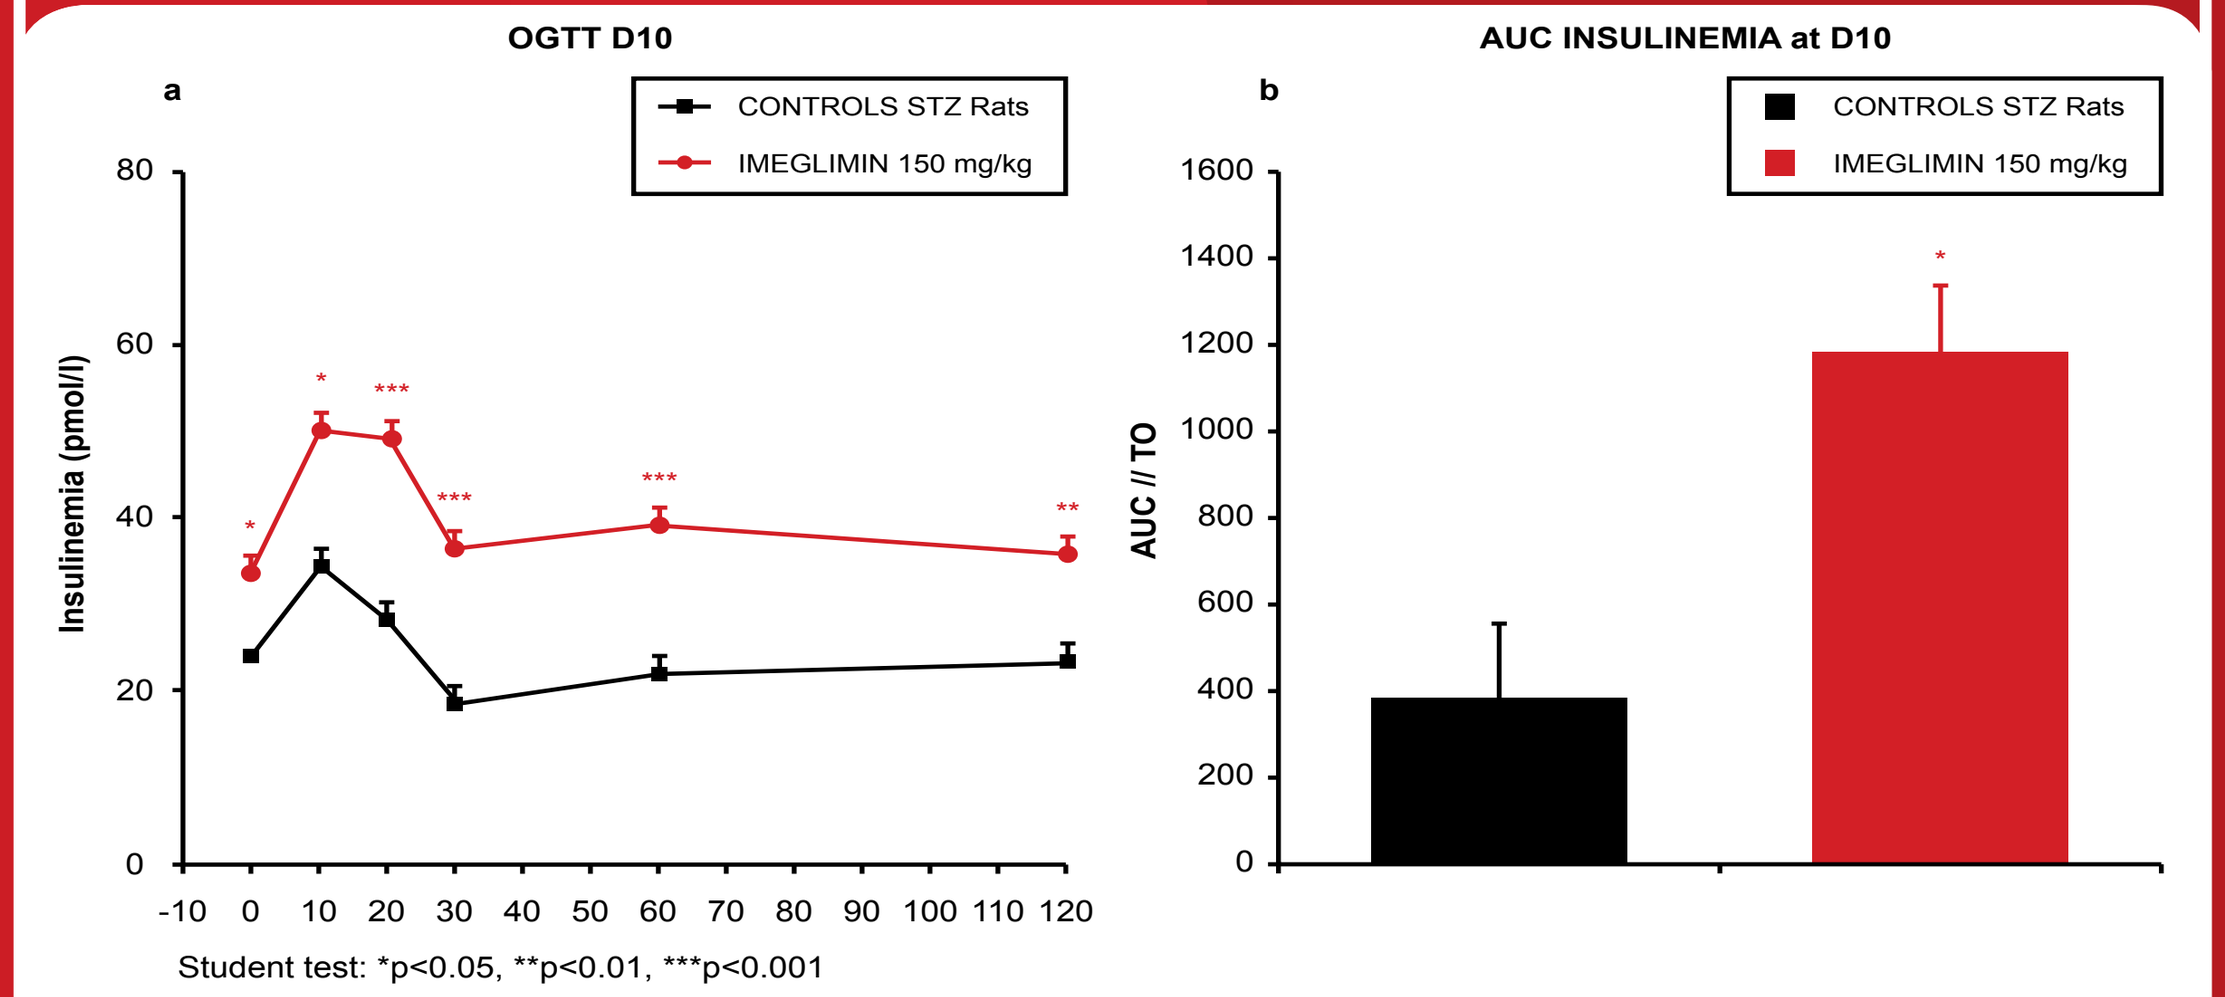
a
OGTT D10
CONTROLS STZ Rats
IMEGLIMIN 150 mg/kg
Insulinemia (pmol/l)
Student test: *p<0.05, **p<0.01, ***p<0.001
b
AUC INSULINEMIA at D10
AUC // TO

Figure 5a and 5b
AUC, area under curve; D10, treatment day 10; OGTT, oral glucose tolerance test; STZ,streptozotocin

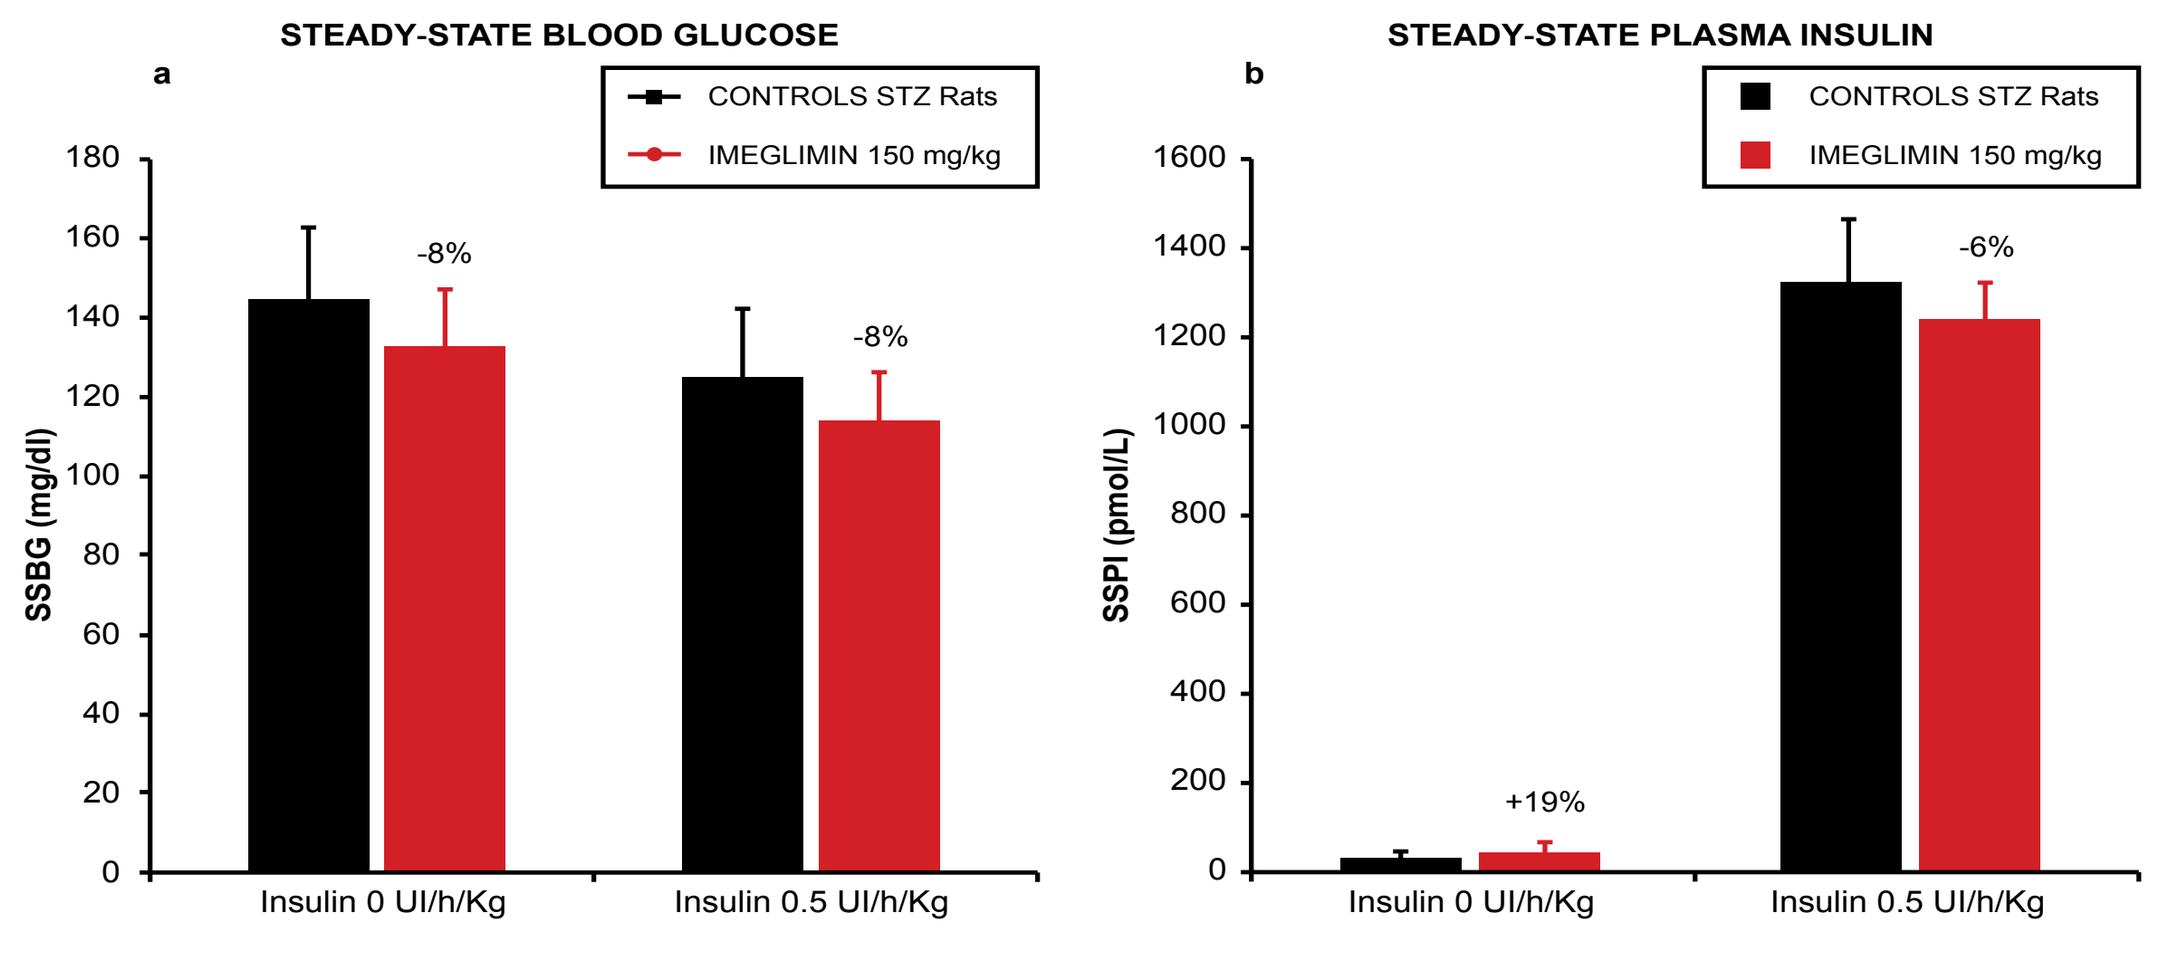
a
STEADY-STATE BLOOD GLUCOSE
CONTROLS STZ Rats
IMEGLIMIN 150 mg/kg
SSBG (mg/dl)
-8%
-8%
Insulin 0 UI/h/Kg
Insulin 0.5 UI/h/Kg
b
STEADY-STATE PLASMA INSULIN
SSPI (pmol/L)
+19%
-6%

Figure 6a and 6b
SSBG, steady-state plasma glucose; SSPI, steady-state plasma insulin; STZ, streptozotocin

Imeglimin decreased GPR and improved hepatic insulin sensitivity during euglycemic hyperinsulinemic clamp

- Under euglycemic hyperinsulinemic clamp conditions, the levels of glucose and insulin were similar in Imeglimin-treated STZ rats infused with insulin as compared with the control STZ rats receiving the same infusion of insulin (Figure 6a and 6b)
- Under these conditions of similar glycemia and insulinemia in both groups, Imeglimin treatment induced a significant increase in the SSGIR required to maintain euglycemia during the clamp, indicating increased overall insulin sensitivity (+209%; p<0.01) (Figure 7)
- Under these conditions, overall GUR was slightly, but not significantly, increased (+23%; p=0.06) (Figure 7)
- In the presence of hyperinsulinemia, Imeglimin treatment was shown to decrease hepatic GPR by 40% (p<0.05) as compared with the controls, indicating an increased hepatic insulin sensitivity (Figure 7)

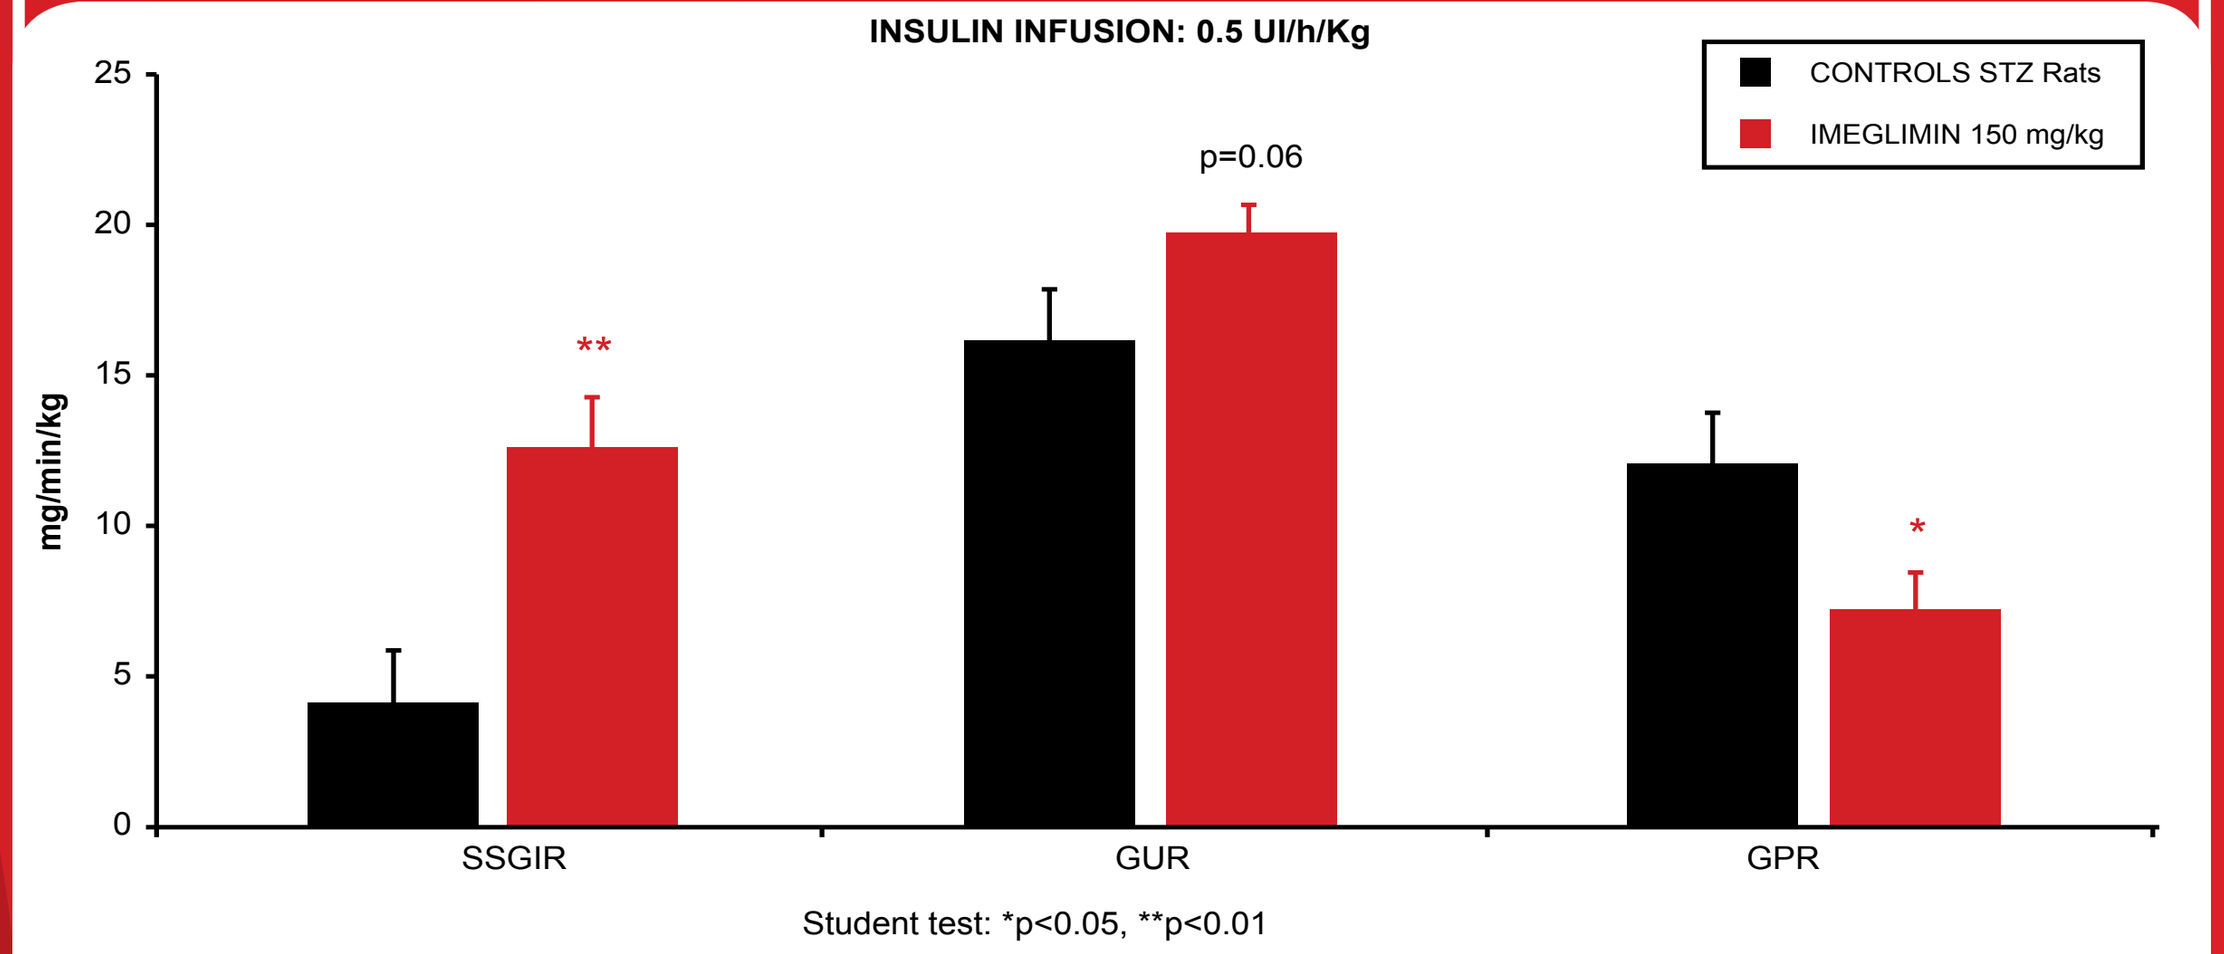
INSULIN INFUSION: 0.5 UI/h/Kg
CONTROLS STZ Rats
IMEGLIMIN 150 mg/kg
mg/min/kg
p=0.06
SSGIR
GUR
GPR
Student test: *p<0.05, **p<0.01

Figure 7
GPR, glucose production rate; GUR, glucose utilization rate; SSGIR, steady-state glucose infusion rate; STZ, streptozotocin

Conclusions

- Imeglimin induced a beneficial effect on glucose tolerance after both acute and chronic administration in overnight-fasted STZ-treated rats
- During the euglycemic hyperinsulinemic clamp, Imeglimin was shown to improve overall insulin sensitivity (SSGIR)
- Imeglimin significantly improved hepatic insulin sensitivity
- Glucose disposal was increased after Imeglimin treatment albeit not significantly
- In addition, Imeglimin was able to maintain basal insulinemia at the same levels as control rats from Day 1 to Day 10; this suggests preservation of β-cell function and/or mass in Imeglimin-treated STZ rats

Abbreviations

AUC, area under curve; GPR, glucose production rate; GSIS, glucose-stimulated insulin secretion; GUR, glucose utilization rates; MOA, mechanism of action; Ra, rate of glucose appearance; Rd, rate of glucose disappearance; SSGIR, steady-state glucose infusion rate; STZ, streptozotocin

Conflicts of Interests

Sophie Hallakou-Bozec and Sebastien Bolze are Poxel employees.

Micheline Kergoat is an employee of Metabrain Research. Metabrain Research performed these experiments.

References

1. Pirags V, et al. *Diabetes Obes Metab* 2012;14:852–858; 2. Vial G, et al. *Diabetes* 2015;64:2254–2264; 3. Fouqueray P, et al. *J Diabetes Metab* 2011;2:126; 4. Pacini G, et al. *Diabetes Obes Metab* 2015;17:541–545

Corresponding author: Sophie Bozec, 259-261 avenue Jean Jaurès, 69007 Lyon, France

Email: sophie.bozec@poxelpharma.com