00081. World Congress on Insulin Resistance, Diabetes & Cardiovascular Disease, 19th–21st November, Los Angeles, California, USA

PXL770 Demonstrates Therapeutic Potential as a New Direct AMP Kinase Activator

Sophie Hallakou-Bozec,[1] Sebastien Bolze,[1] Marc Foretz[2]

[1]Poxel SA, Lyon, France; [2]Institut Cochin - INSERM U1016 - CNRS UMR8104 - Université Descartes Paris Département Endocrinologie, France

Abstract

AMPK is a heterotrimeric kinase playing a major role in regulating cellular energy balance. AMPK has raised widespread interest as a potential therapeutic target for metabolic diseases. Direct AMPK activation is expected to decrease insulin resistance, lipid disorders and to improve glycemic control. PXL770 is a small molecule, shown to directly activate all AMPK holoenzymes tested, with a higher potency for β1-containing isotypes. PXL770 activates AMPK allosterically and protects AMPK against dephosphorylation, as demonstrated in several recombinant AMPK isoforms. PXL770 binds to AMPK differently from the natural AMPK ligand, AMP, and its binding site involves the CBM region in the β subunit. Cell-based assay confirmed that PXL770 inhibits hepatic *de novo* lipogenesis in an AMPK-dependent manner. PXL770 exhibits good oral availability and dose linear pharmacokinetic profile. In an obese type 2 diabetic rodent model (ob/ob mice), orally administered PXL770 during 6 weeks significantly and dose dependently improves glucose tolerance without an increase in insulin levels suggesting an insulin sensitizing effect. PXL770 significantly improved HbA_{1c}, decreased plasma triglycerides as well as liver weight and triglycerides content showing an improvement in liver steatosis. In the same model, we showed that PXL770 induced a significant increase of phosphorylated AMPK levels in both liver and muscle, demonstrating *in vivo* target engagement. In conclusion, PXL770 is a novel direct AMPK activator that improves both glycemic control and lipid profile, the two main cardiovascular risk factors, and potentially could be a new oral agent for the treatment of type 2 diabetes and dyslipidemia.

Background

- Ectopic accumulation of hepatic lipids is linked to the development of hepatic insulin resistance and type 2 diabetes[1–3]
- Therapies directed at improving insulin resistance may slow or even arrest the progression of type 2 diabetes[4]
- Activated AMPK switches cells from an anabolic to a catabolic state, shutting down the ATP-consuming pathways and restoring energy balance via modulation of gene expression[5]
- Because of its key role in energy metabolism, AMPK is an emerging therapeutic target for the treatment of type 2 diabetes and cardiovascular complications

Objectives

- The aim of this study was to investigate the direct effect of PXL770 on AMPK activation, hepatic lipogenesis *in vitro*, and glycemic control and lipid profile *in vivo* in ob/ob mice

Research Design and Methods

Expression of recombinant proteins and AMPK assays

- All AMPK complexes were expressed in *E. coli* using tricistronic vectors
- AMPK activity was determined by phosphorylation of the SAMS peptide in the presence or absence of varying PXL770 concentrations
- For dephosphorylation experiments, recombinant AMPK complex (0.5–1 µg) was incubated in the presence or absence of recombinant PP2C (10–20 ng) and 2.5 mM $MgCl_2$, in the presence or absence of varying concentrations of PXL770 for 15 minutes at 37°C. Dephosphorylation step was stopped and AMPK activity was measured using the SAMS peptide assay

Analysis of lipogenesis in murine wild-type and liver-specific AMPKα1/α2 KO hepatocytes

- Primary hepatocytes were isolated from fed adult male mice and were maintained in M199 medium in the presence of either 5 mM glucose or 25 mM glucose plus 100 nM insulin, and various concentrations of PXL770 (10–100 µM), or 50 µM cerulenin (fatty acid synthase inhibitor)
- After 20 hours, the culture medium was removed and cell triglyceride (TG) content was assayed using a commercial kit. TG contents were normalized to protein content measured in adjacent wells by using the bicinchoninic acid assay

Glycemic control, triglyceride content and P-AMPK levels in vivo *in ob/ob mice*

- Male ob/ob mice and ob+ mice (n=10/group) were purchased from CERJ (53940 Le Genest Saint Isle, France). All animals were housed in a temperature-controlled environment on a 12-hour light-dark cycle (light on at 7:00 am) and had access to food and water *ad libitum*
- PXL770 was administered twice daily for 5 weeks via the oral route at 25 mg/kg, 50 mg/kg or 100 mg/kg
- At day 36, plasma glucose levels, HbA_{1c} and triglycerides were measured
- Liver samples from mice were collected and homogenized in tubes containing lysis buffer and centrifuged; pellets were used for measuring triglyceride content using a commercial kit
- In a separate experiment, PXL770 was administered to ob/ob mice for 5 days at 75 mg/kg twice daily. Animals were sacrificed and liver and muscle tissues were collected for P-AMPK measurements. P-AMPK levels were determined using an ELISA kit

Results

PXL770 is a potent direct activator of AMPK in vitro

- PXL770 allosterically activates and/or protects α1β2γ1, α2β2γ3, α1β1γ1, α2β1γ1, and α2β2γ1 complexes against dephosphorylation to different extents and with different potency (Table 1)

Table 1: Mechanisms of direct AMPK activation by PXL770

	AMPK activity	
Isoforms tested	EC50 allosteric activation	EC50 dephosphorylation protection
α1β2γ1	No effect	~4 µM
α2β2γ3	~2.2 µM	*Not tested*
α2β2γ1	~1 µM	~1.3 µM
α2β1γ1	~40–80 nM	~700 nM
α1β1γ1	~30 nM	~250 nM

AMPK, 5' adenosine monophosphate-activated protein kinase; EC, effective concentration

- Deletion of the CBM (N-terminal 185 residues) or within the CBM (mutation of serine 108 to alanine) from β1 abolishes protection against dephosphorylation by PXL770 meaning that binding involves the CBM region of AMPK (Figure 1)

Figure 1: Intact CBM region in the β subunit is necessary for the binding of PXL770

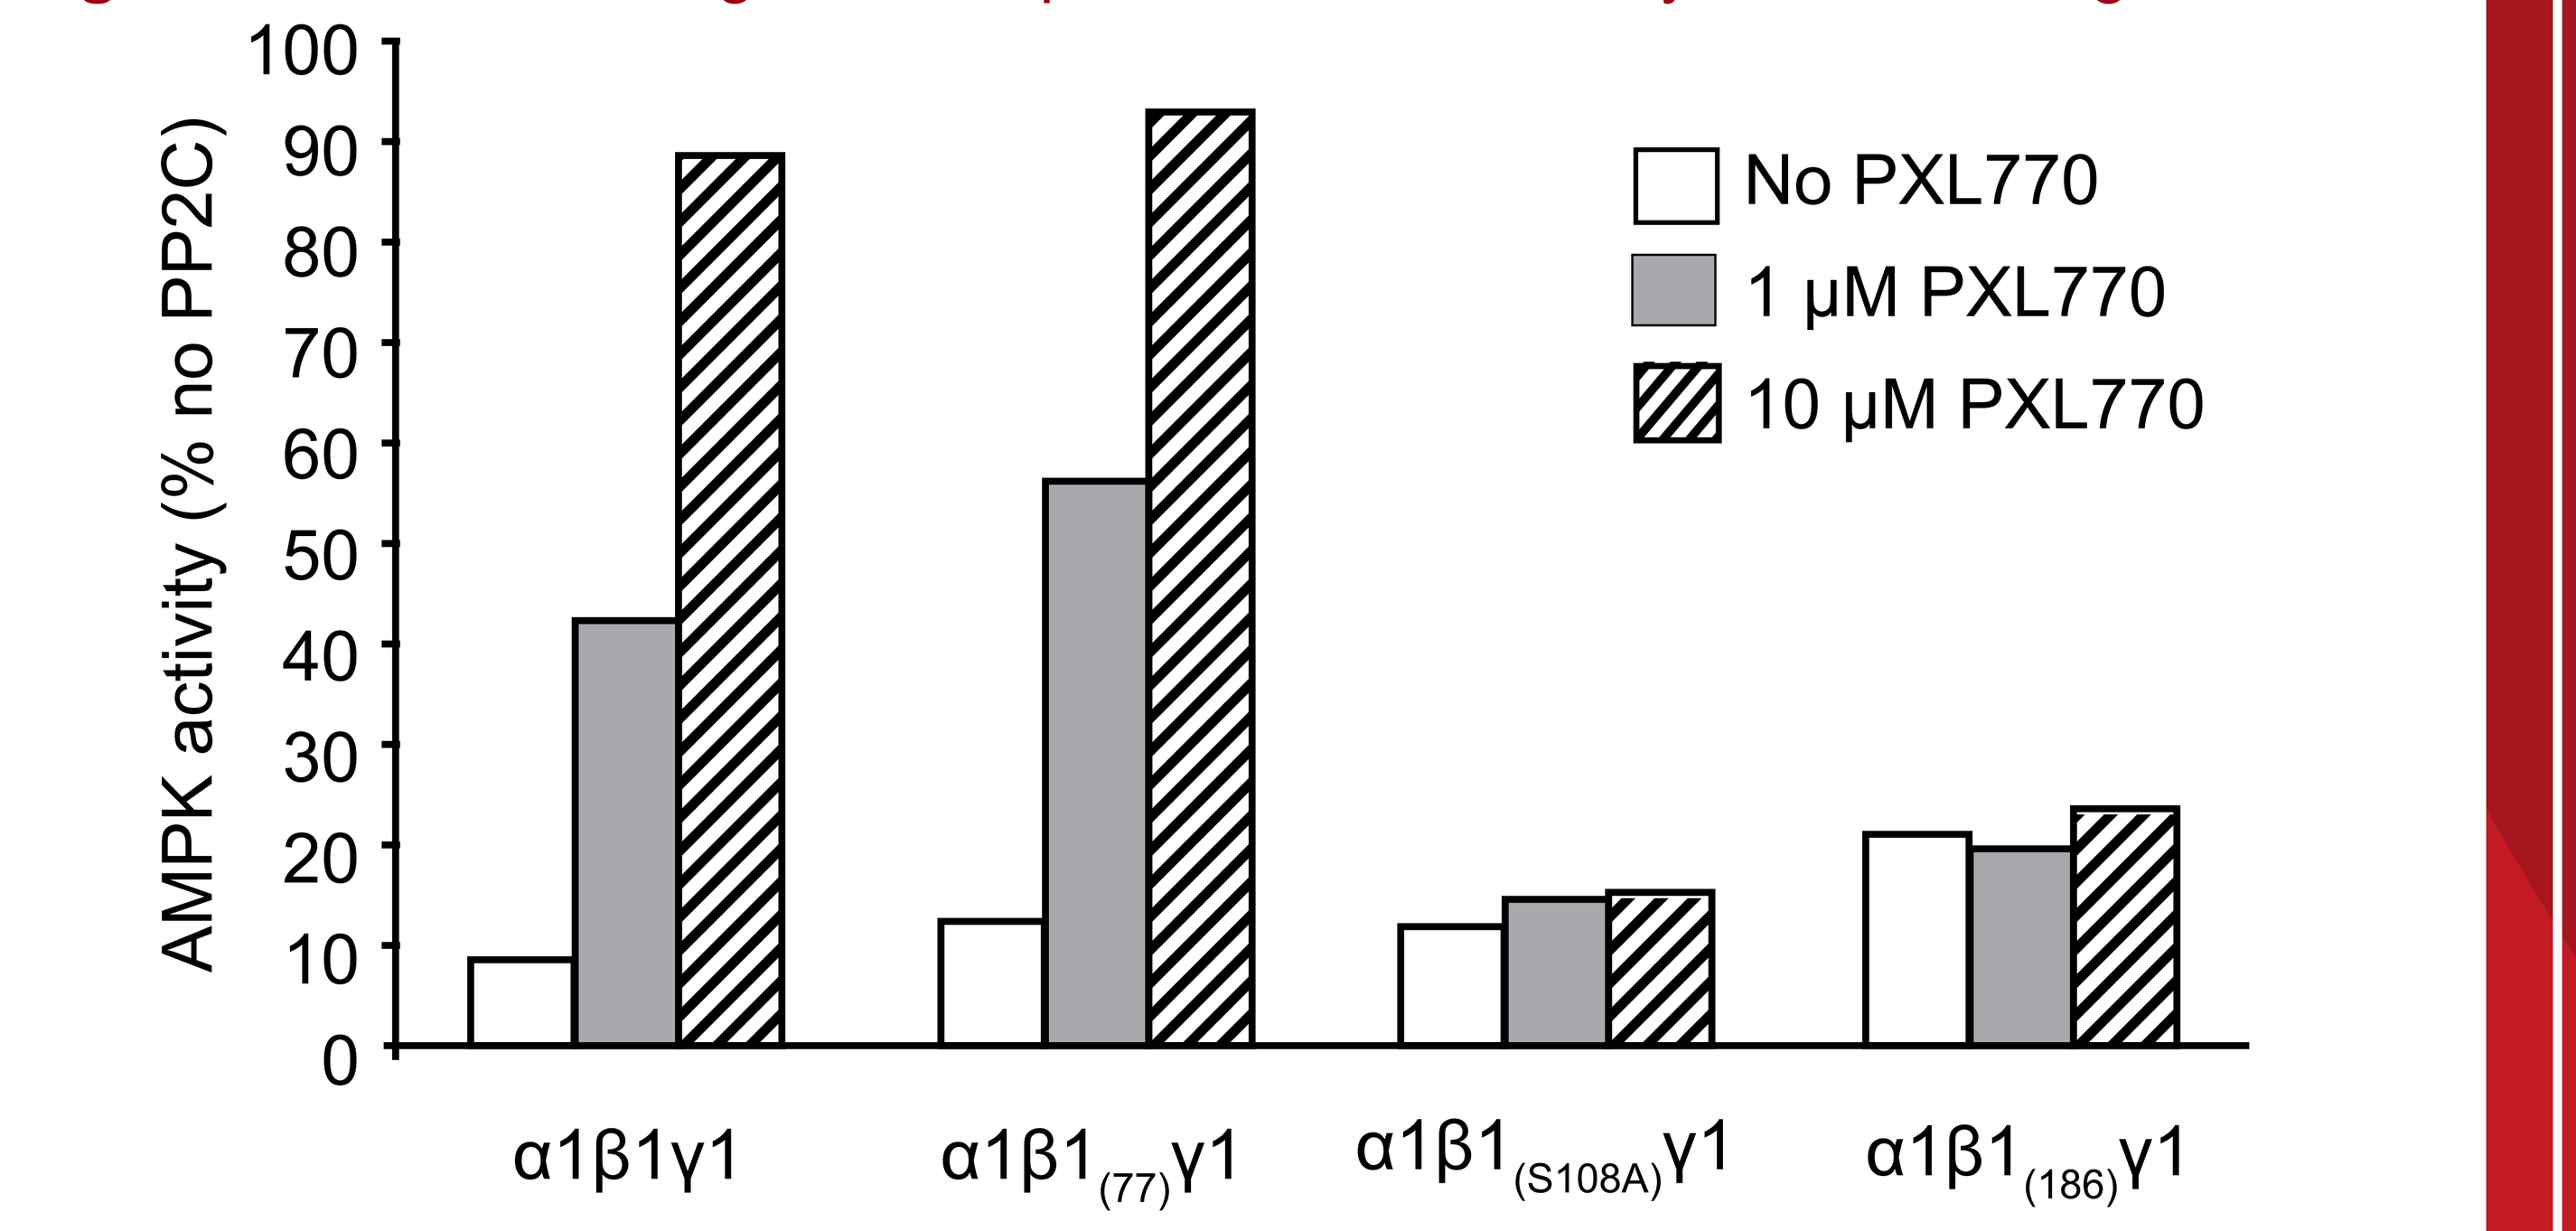
AMPK activity (% no PP2C)
100
90
80
70
60
50
40
30
20
10
0
No PXL770
1 µM PXL770
10 µM PXL770
α1β1γ1
α1β1(77)γ1
α1β1(S108A)γ1
α1β1(186)γ1

CBM, carbohydrate-binding module; PP2C, protein phosphatase 2c

- PXL770 did not compete for binding of nucleotide to the γ1 subunit, suggesting that the compound does not bind to the nucleotide binding sites of AMPK (data not shown)

PXL770 inhibits de novo *lipogenesis in vitro via AMPK activation*

- TG accumulation was increased in both wild-type and AMPKα1α2-null hepatocytes after incubation with 25 mM glucose and 100 nM insulin, but was inhibited in the presence of cerulenin, indicating that this increase was dependent on lipogenesis. In wild-type hepatocytes, TG accumulation was decreased in a dose-dependent manner upon treatment with increasing concentrations of PXL770; this was not the case in AMPKα1α2-null hepatocytes, demonstrating the AMPK dependency (Figure 2a and b)

Figure 2a and b: PXL770 reduced hepatocyte lipogenesis via AMPK activation

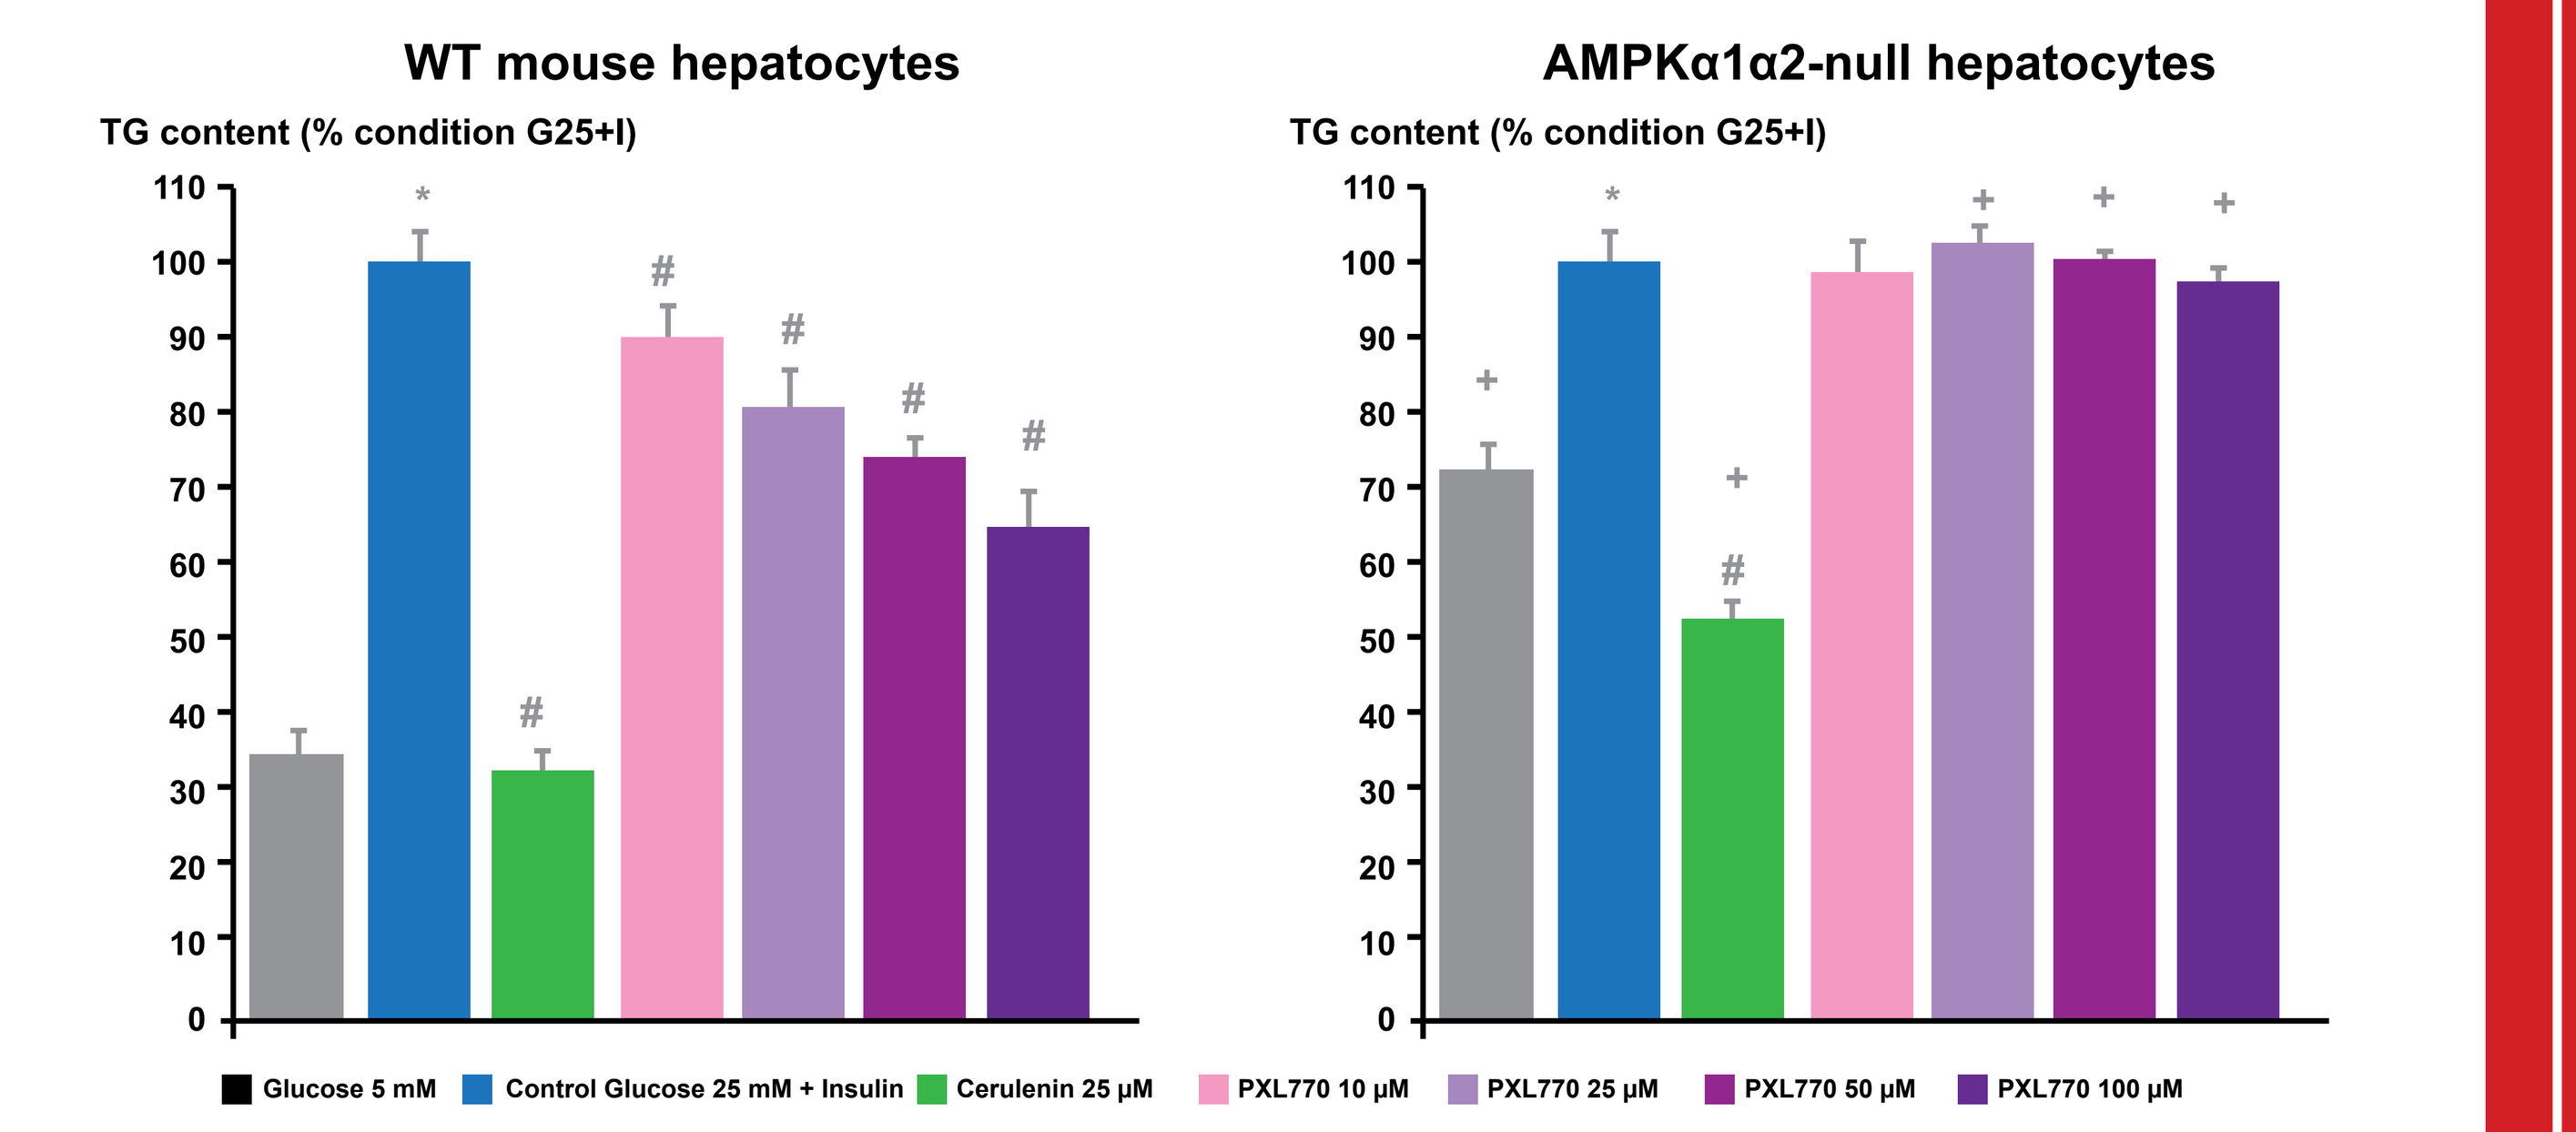
WT mouse hepatocytes
AMPKα1α2-null hepatocytes
TG content (% condition G25+I)
Glucose 5 mM
Control Glucose 25 mM + Insulin
Cerulenin 25 µM
PXL770 10 µM
PXL770 25 µM
PXL770 50 µM
PXL770 100 µM

*p<0.05 compared with hepatocytes of the same genotype in G5 mM
#p<0.05 compared with hepatocytes of the same genotype in G25 mM+insulin
+p<0.05 compared with wild-type hepatocytes treated the same condition
AMPK, 5' adenosine monophosphate-activated protein kinase; G, glucose; I, insulin; TG, triglyceride; WT, wild-type

- Similar results were observed in human primary hepatocytes (IC50 ~3 µM vs ~2.8 µM in murine cells [data not shown])

PXL770 improves glycemic control and lipid profile in ob/ob mice

- PXL770 improved glucose tolerance, with a marked decrease in incremental AUC glucose of –27% and –35% at 50 and 100 mg/kg bid, respectively. After treatment with 100 mg/kg bid, glucose tolerance was normalized, reaching that of non-diabetic mice (Figure 3a)

Figure 3a and b: PXL770 improved glycemic control in ob/ob mice

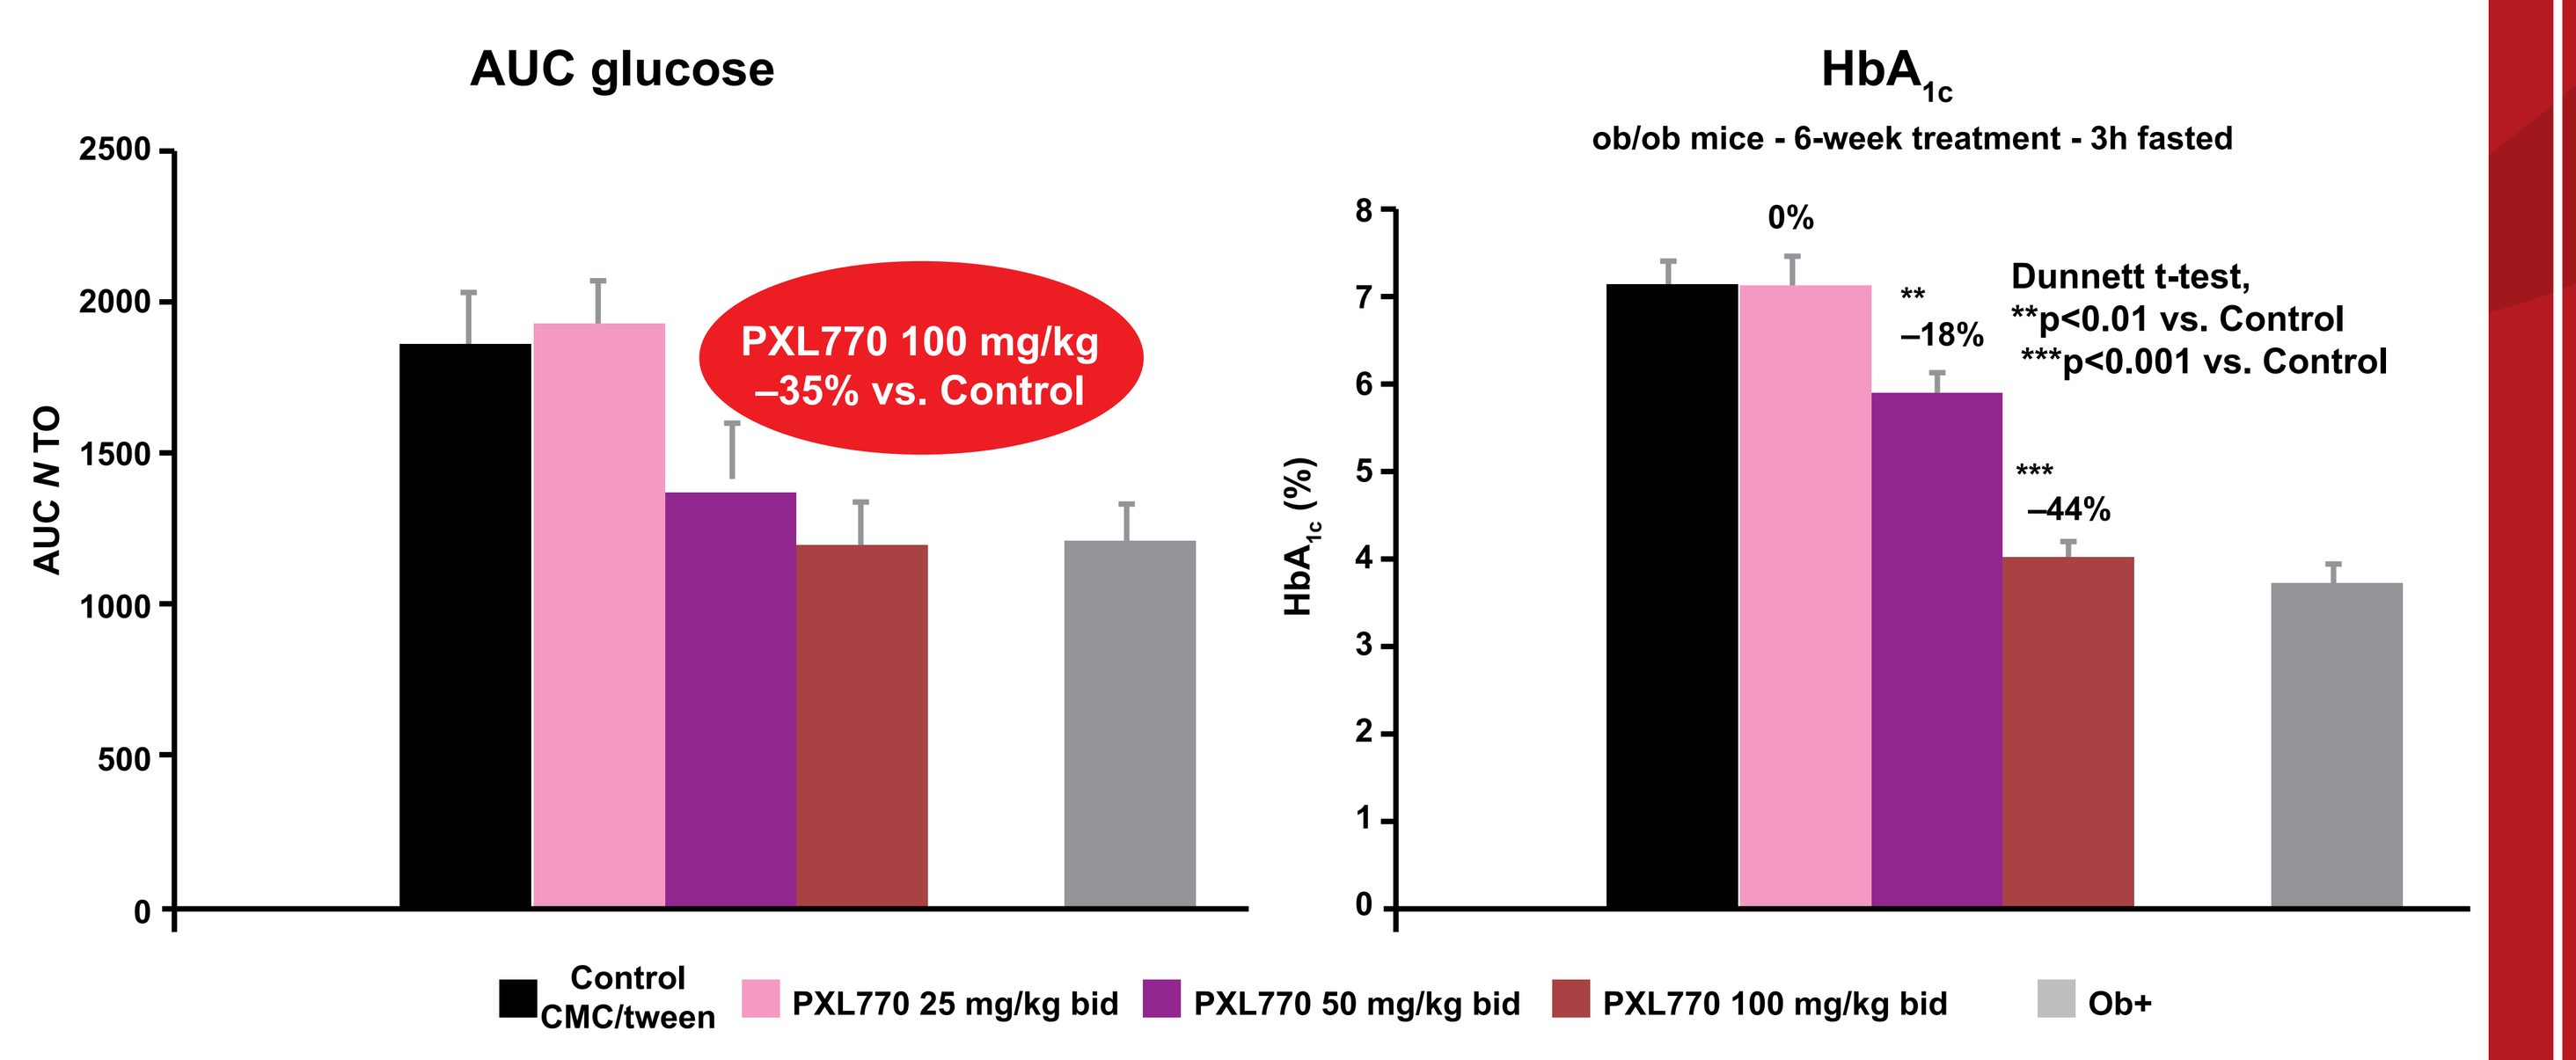
AUC glucose
AUC N TO
PXL770 100 mg/kg
–35% vs. Control
HbA1c
ob/ob mice - 6-week treatment - 3h fasted
HbA1c (%)
0%
–18%
–44%
Dunnett t-test,
**p<0.01 vs. Control
***p<0.001 vs. Control
Control CMC/tween
PXL770 25 mg/kg bid
PXL770 50 mg/kg bid
PXL770 100 mg/kg bid
Ob+

AUC, area under curve; bid , twice daily; CMC, carboxymethylcellulose; HbA_{1c}, glycated hemoglobin; ob/ob, obese; ob+, normal mouse

- A significant dose-dependent effect of PXL770 was observed with respect to HbA_{1c}, with the 100 mg/kg dose resulting in a near-complete normalization of HbA_{1c} level relative to ob+ normal mouse (Figure 3b)
- Treatment with PXL770 also resulted in marked decreases in plasma TGs (–19% NS, –40% [p<0.01], and –43% [p<0.001] at 25 mg/kg, 50 mg/kg, and 100 mg/kg, respectively) and hepatic TG content (Figure 4a and b)

Figure 4a and b: Treatment with PXL770 was associated with marked decreases in both plasma and hepatic triglyceride content in ob/ob mice

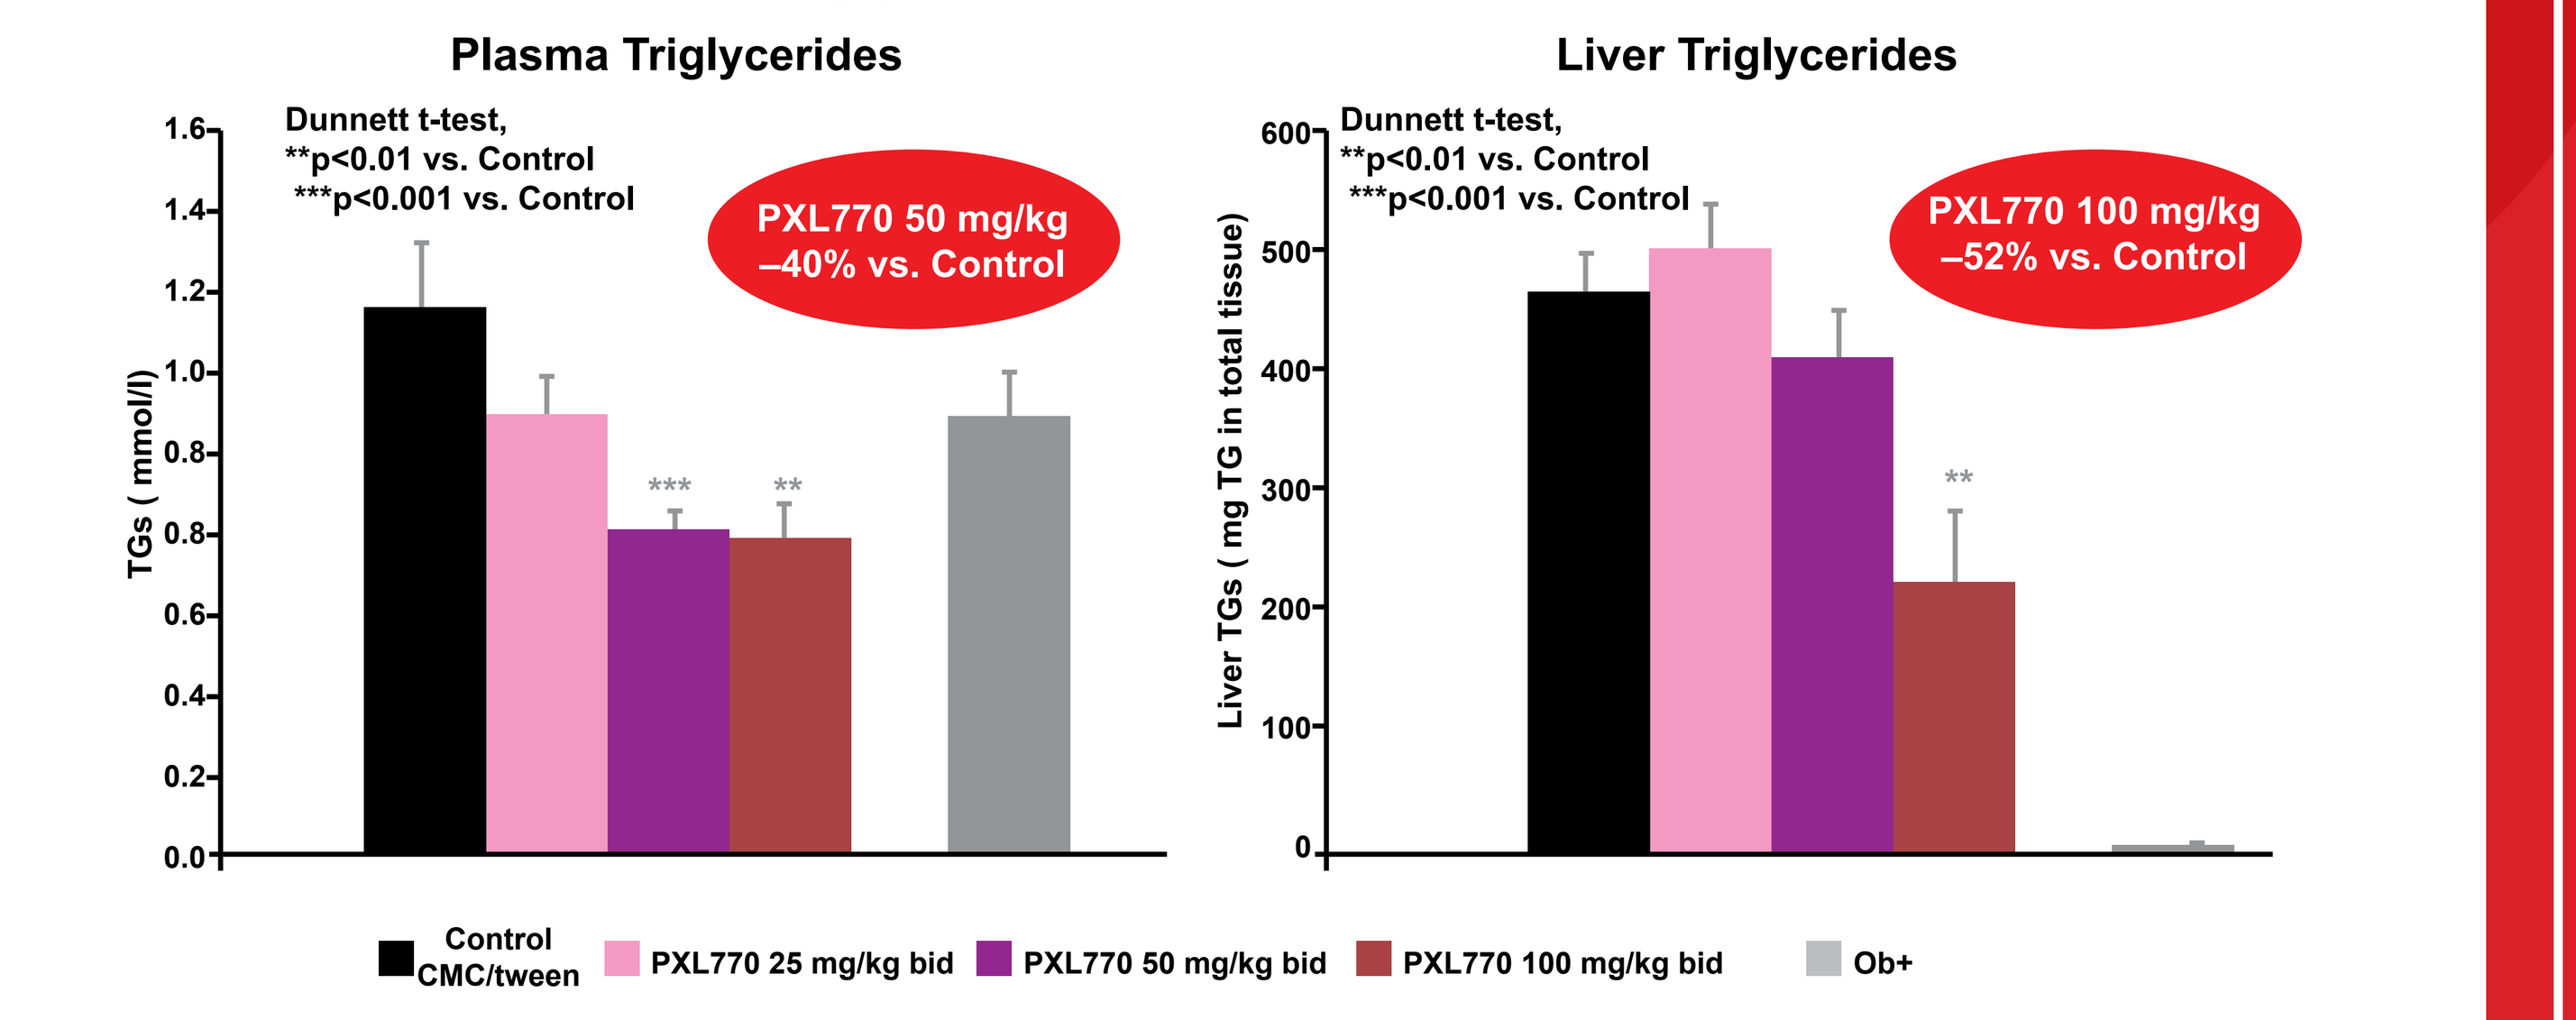
Plasma Triglycerides
Liver Triglycerides
TGs (mmol/l)
Liver TGs (mg TG in total tissue)
Dunnett t-test,
**p<0.01 vs. Control
***p<0.001 vs. Control
PXL770 50 mg/kg
–40% vs. Control
PXL770 100 mg/kg
–52% vs. Control
Control CMC/tween
PXL770 25 mg/kg bid
PXL770 50 mg/kg bid
PXL770 100 mg/kg bid
Ob+

bid, twice daily; CMC, carboxymethylcellulose; ob/ob, obese; ob+, normal mouse; TG, triglyceride

- After 5 days of treatment with 75 mg/kg bid, PXL770 significantly reduced plasma glucose by 40% (data not shown) with a concomitant and significant increase in liver and muscle P-AMPK level (Figure 5a and b)

Figure 5a and b: PXL770 significantly increased P-AMPK levels in liver and muscle tissue of ob/ob mice

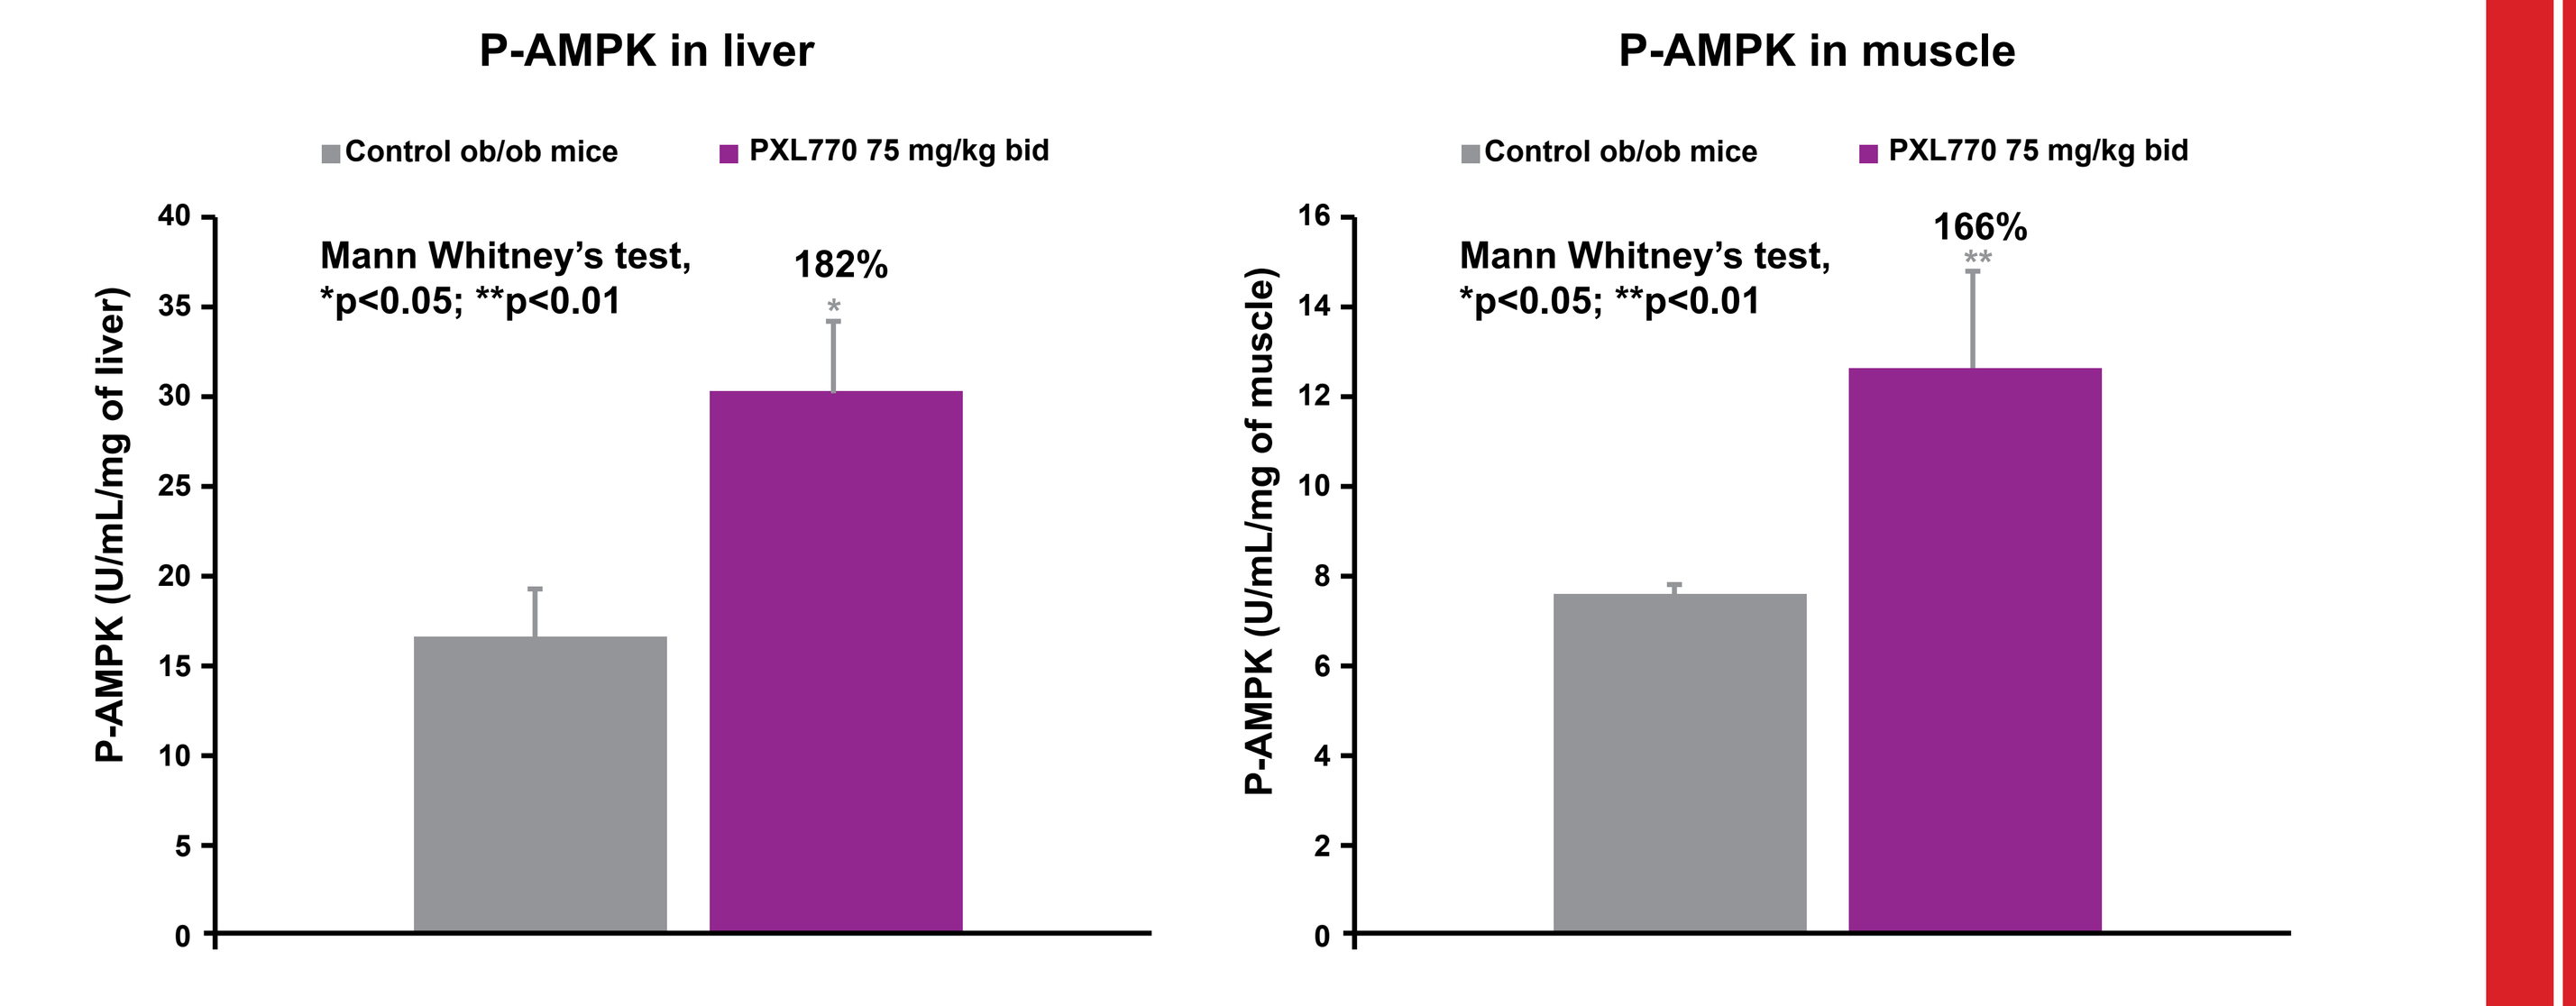
P-AMPK in liver
P-AMPK in muscle
Control ob/ob mice
PXL770 75 mg/kg bid
Mann Whitney's test,
*p<0.05; **p<0.01
182%
166%
P-AMPK (U/mL/mg of liver)
P-AMPK (U/mL/mg of muscle)

bid, twice daily; ob/ob, obese; P-AMPK, phosphorylated 5' adenosine monophosphate-activated protein kinase

- PXL770 is orally bioavailable and exhibits a good pharmacokinetic profile for further development (data not shown)

Conclusion

- In this series of experiments, PXL770, a novel drug candidate, has been shown to potently and directly activate AMPK, inhibit de novo lipogenesis *in vitro*, with a concomitant decrease in liver steatosis, and improved glycemic control and lipid profile in ob/ob mice
- PXL770 exhibits oral bioavailability, which makes it a promising candidate for the treatment of patients with type 2 diabetes and dyslipidemia, who are at a higher risk of cardiovascular complications
- PXL770 is ready for phase 1

Acknowledgments

We thank Dr David Carling and Dr Steve Gamblin for their contributions to these data.

References

1. Jornayvaz FR, et al. *Cell Metab* 2012;15:574–584; 2. Cusi K. *Gastroenterology* 2012;142:711–725; 3. Birkenfeld AL, et al. *Hepatology* 2014;59:713–23; 4. Tolman KG, et al. *Ther Clin Risk Manag* 2007;3:1153–1163; 5. Violett B, et al. *Acta Physiol* (Oxf) 2009;196:81–98

Presenting author: Sophie Bozec, 259/261 avenue Jean Jaurès F-69007 Lyon
sophie.bozec@poxelpharma.com