PXL770, a new direct AMP Kinase activator, demonstrates promising effects for treatment of non-alcoholic steatohepatitis

Pascale Gluais-Dagorn[1], Pascale Fouqueray[1], Sebastien Bolze[1], Sophie Hallakou-Bozec[1]

[1]Poxel, 259-261 avenue Jean Jaurès, 69007 Lyon, France

Background

- Nonalcoholic fatty liver disease (NAFLD) is the most prevalent hepatic pathology in the Western world and nonalcoholic steatohepatitis (NASH) is estimated to occur in 10-30% of patients with NAFLD
- The AMP-activated protein kinase (AMPK) is a central regulator of multiple metabolic pathways and may have therapeutic importances for treating NAFLD
- PXL770 is a new direct AMPK activator. PXL770 is currently assessed in multiple ascending dose phase 1 study in healthy subjects. PXL770 has been shown to decrease de novo lipogenesis and to broadly improve metabolic profile in various rodent models suggesting that it could play an important role in the management of patients with NAFLD/NASH

Objective

The aim of this study was to assess the effects of PXL770 in a diet induced obesity NASH mouse model

Methods and Design

Male C57BL/6J were fed a diet high in trans fat (40%), fructose (20%) and cholesterol (2%) for a total of 41 weeks. After the diet induction period, a liver biopsy was collected to confirm steatosis and fibrosis status and to stratify the mice into diet induced obesity (DIO)-NASH groups treated per os for 8 weeks (n=12/group) with vehicle, PXL770 35 or 75 mg/kg twice daily or elafibranor (30 mg/kg daily). NAFLD activity score (NAS) and fibrosis score were determined at baseline and at the end of the treatment period based on Hematoxylin Eosin (H&E) stain and Sirius Red respectively. Plasma liver enzymes and lipids as well as liver triglycerides and liver collagen genes expression were assessed

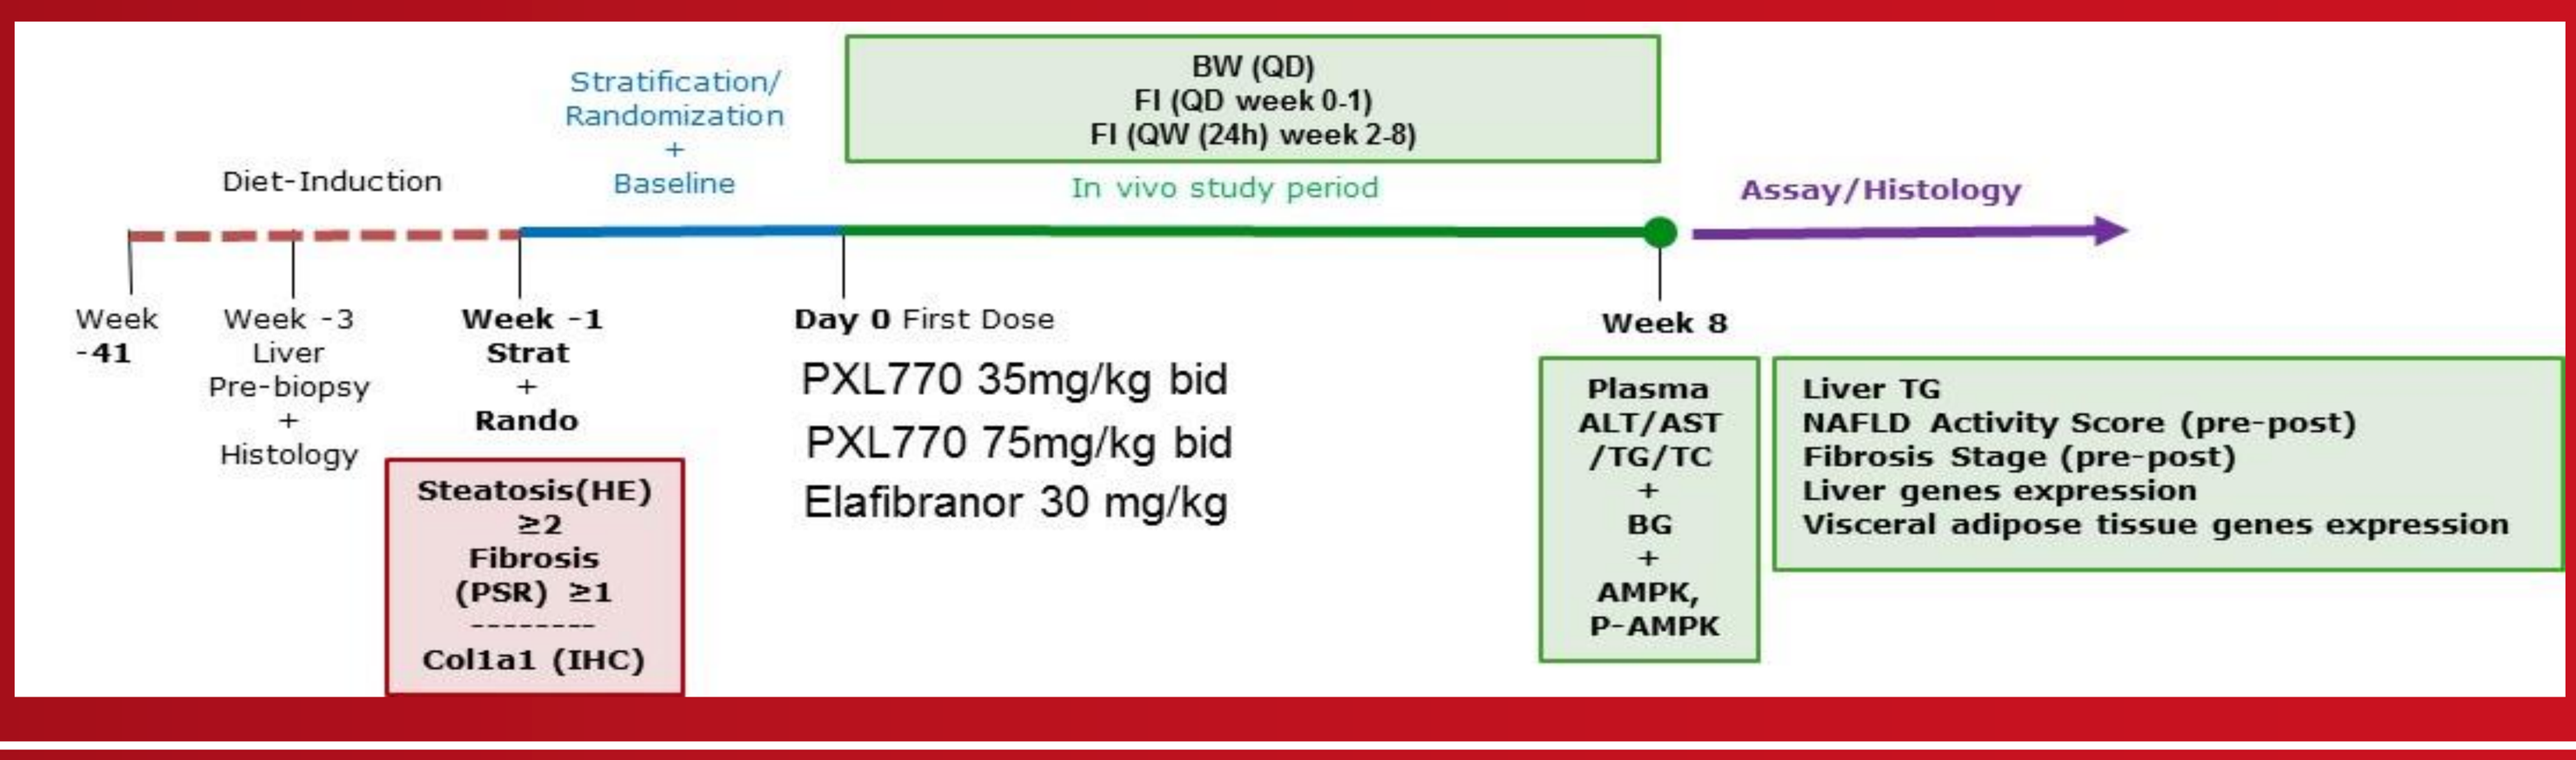
Stratification/ Randomization + Baseline
BW (QD)
FI (QD week 0-1)
FI (QW (24h) week 2-8)
Diet-Induction
In vivo study period
Assay/Histology
Week -41
Week -3 Liver Pre-biopsy + Histology
Week -1 Strat + Rando
Steatosis(HE) ≥2 Fibrosis (PSR) ≥1 -------- Col1a1 (IHC)
Day 0 First Dose
PXL770 35mg/kg bid
PXL770 75mg/kg bid
Elafibranor 30 mg/kg
Week 8
Plasma ALT/AST /TG/TC + BG + AMPK, P-AMPK
Liver TG
NAFLD Activity Score (pre-post)
Fibrosis Stage (pre-post)
Liver genes expression
Visceral adipose tissue genes expression

Results

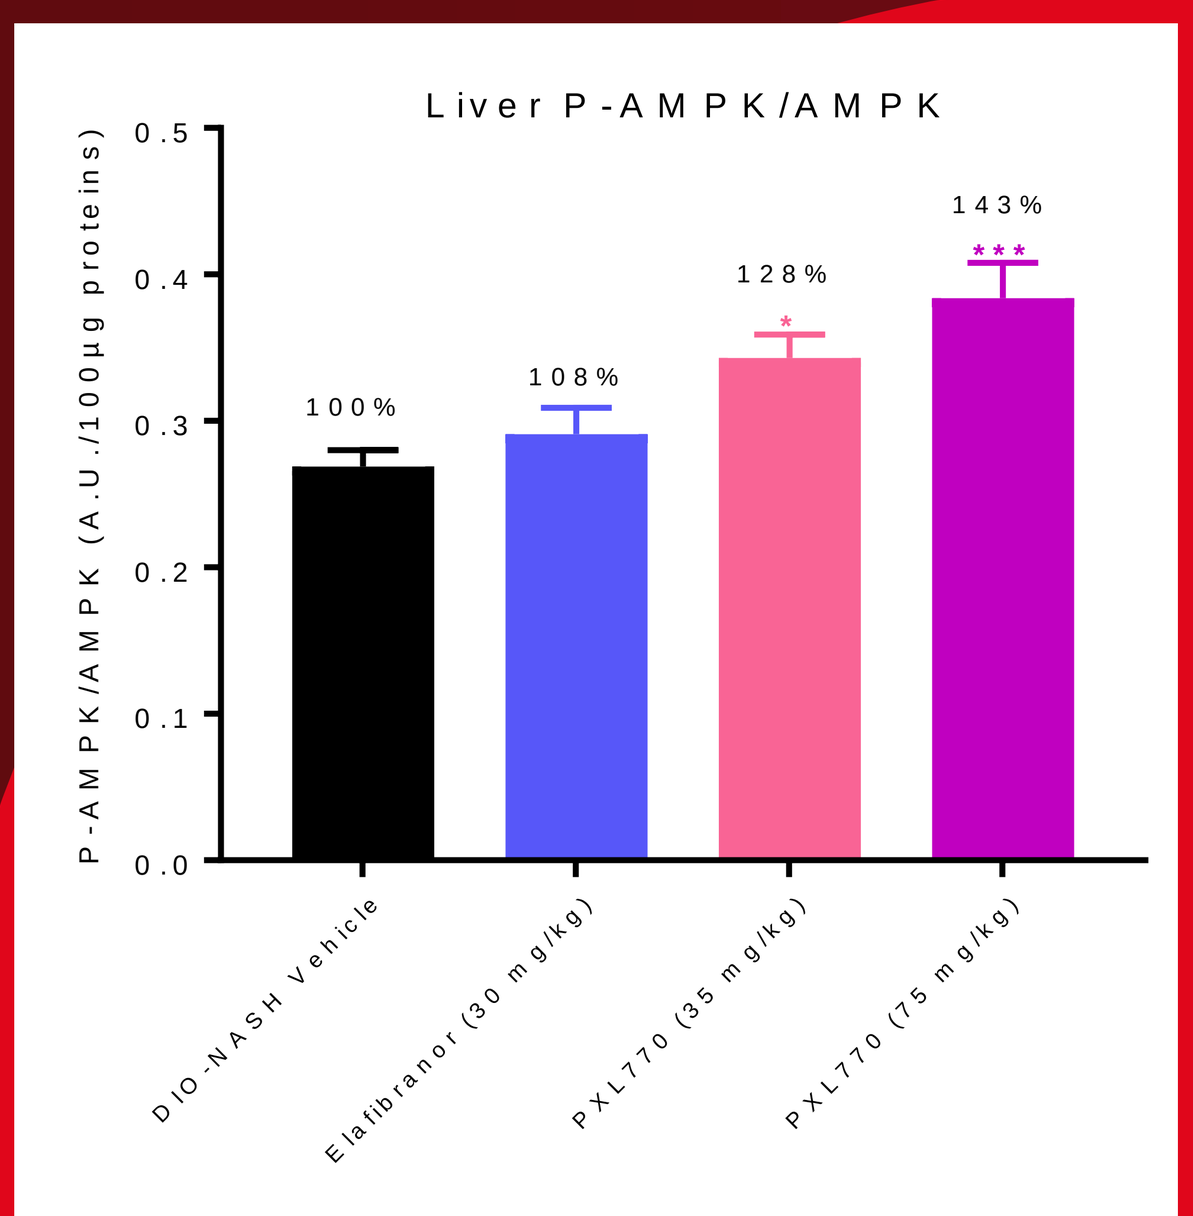
Liver P-AMPK/AMPK
P-AMPK/AMPK (A.U./100µg proteins)
100%
108%
128%
143%
DIO-NASH Vehicle
Elafibranor (30 mg/kg)
PXL770 (35 mg/kg)
PXL770 (75 mg/kg)

Figure 1 - PXL770 increases AMPK phosphorylation in liver tissue

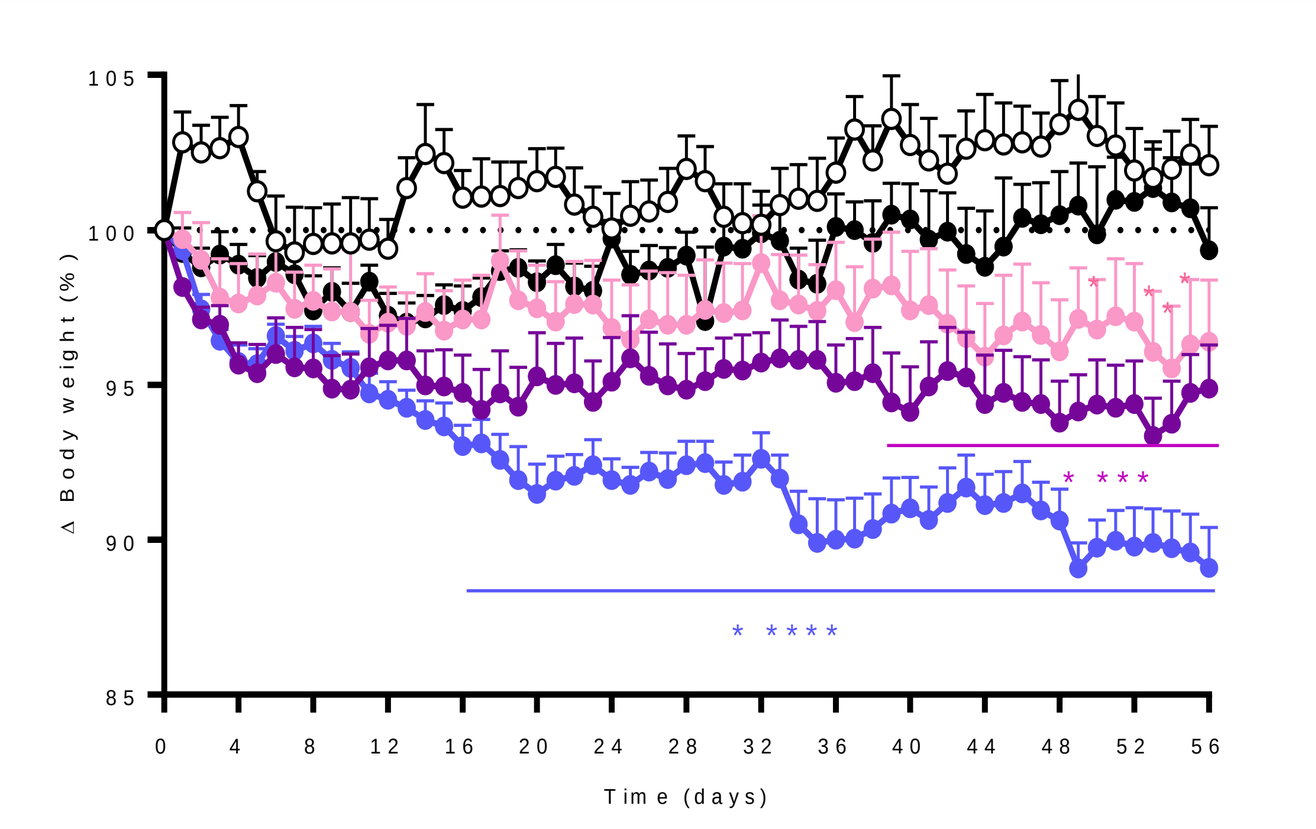
Δ Body weight (%)
Time (days)

*p<0.05, **P<0.01 vs. DIO-NASH Vehicle

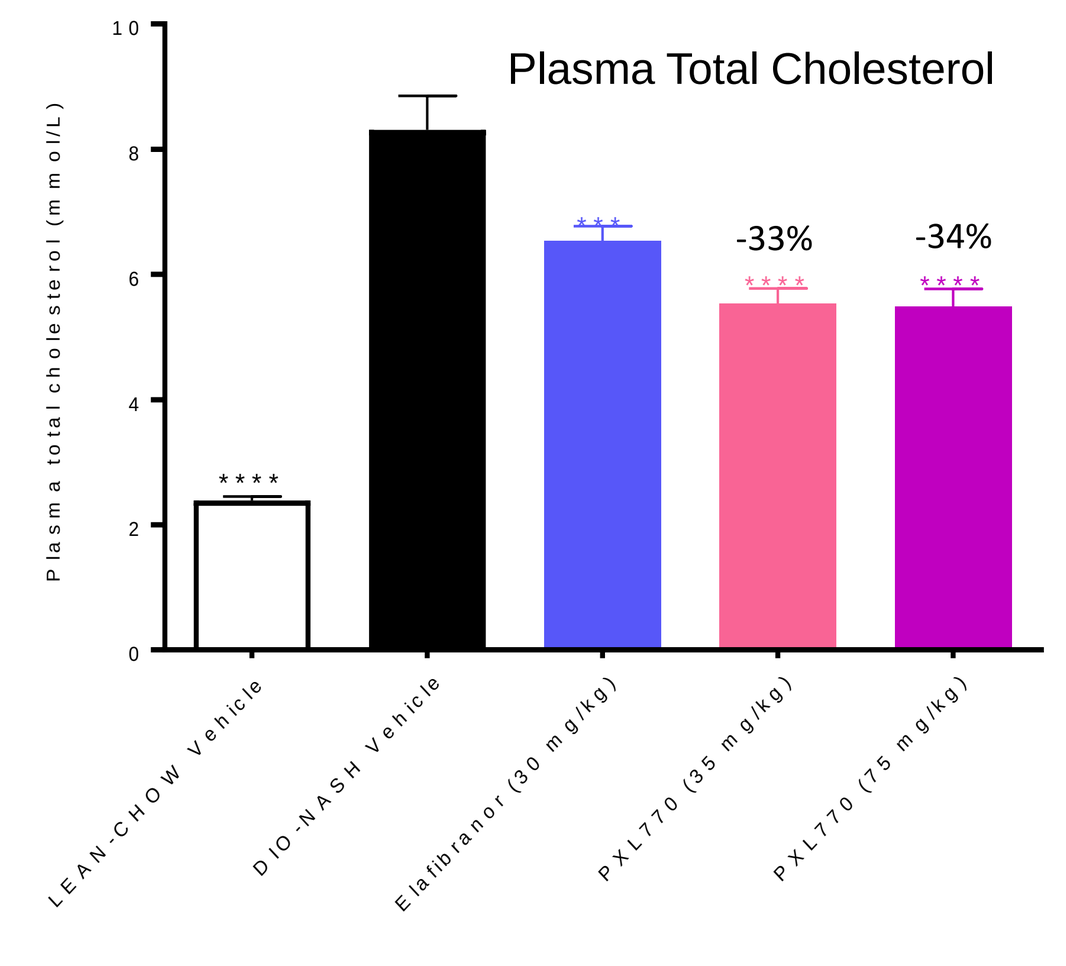
Plasma Total Cholesterol
Plasma total cholesterol (mmol/L)
-33%
-34%
LEAN-CHOW Vehicle
DIO-NASH Vehicle
Elafibranor (30 mg/kg)
PXL770 (35 mg/kg)
PXL770 (75 mg/kg)

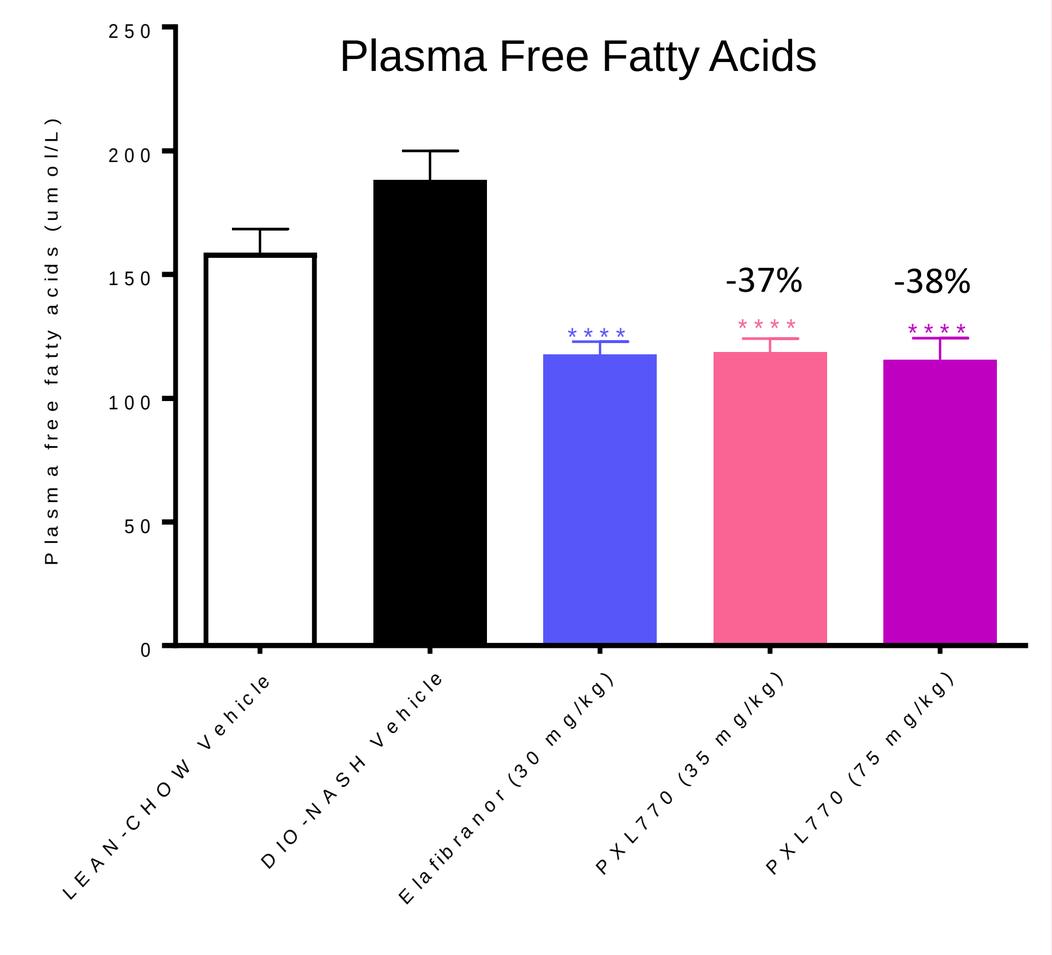
Plasma Free Fatty Acids
Plasma free fatty acids (umol/L)
-37%
-38%
LEAN-CHOW Vehicle
DIO-NASH Vehicle
Elafibranor (30 mg/kg)
PXL770 (35 mg/kg)
PXL770 (75 mg/kg)

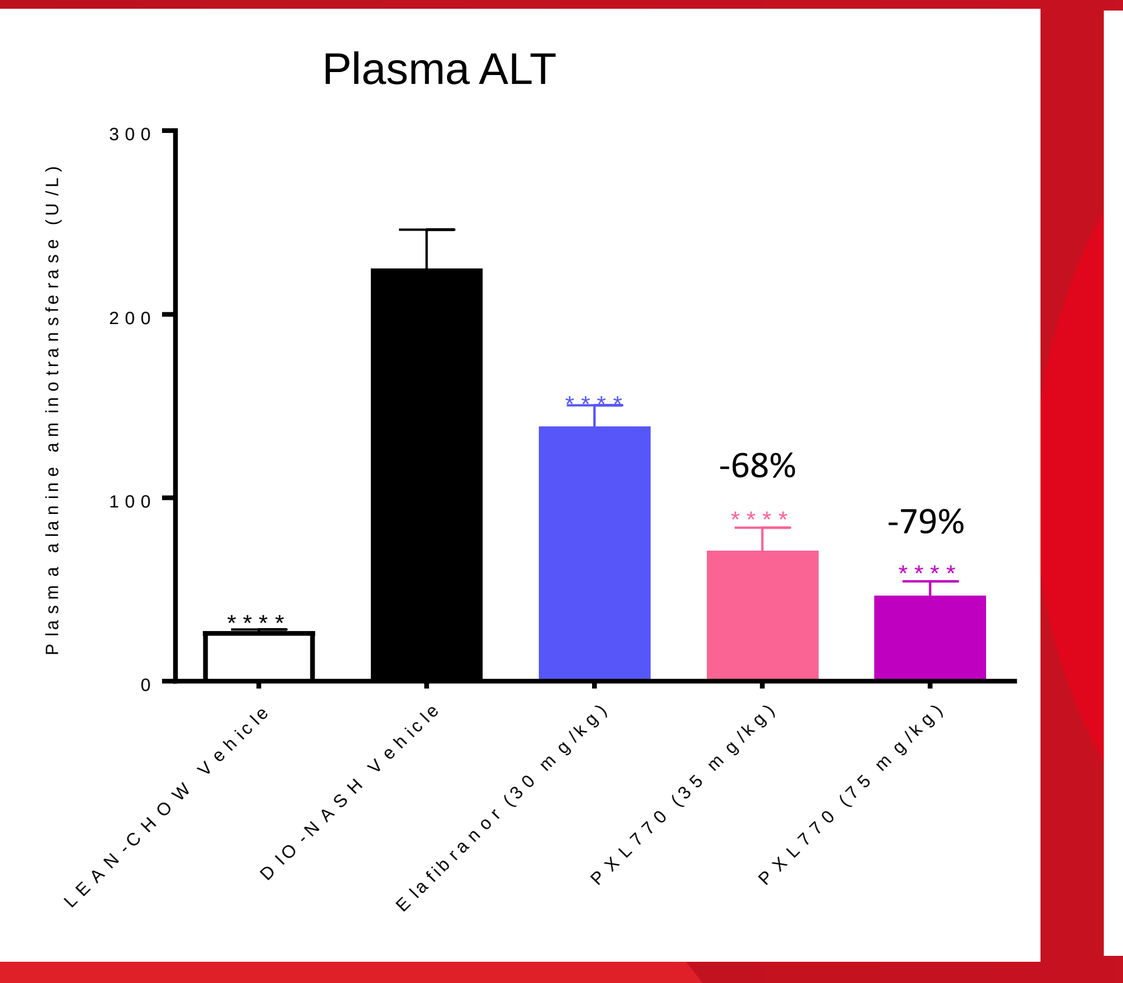
Plasma ALT
Plasma alanine aminotransferase (U/L)
-68%
-79%
LEAN-CHOW Vehicle
DIO-NASH Vehicle
Elafibranor (30 mg/kg)
PXL770 (35 mg/kg)
PXL770 (75 mg/kg)

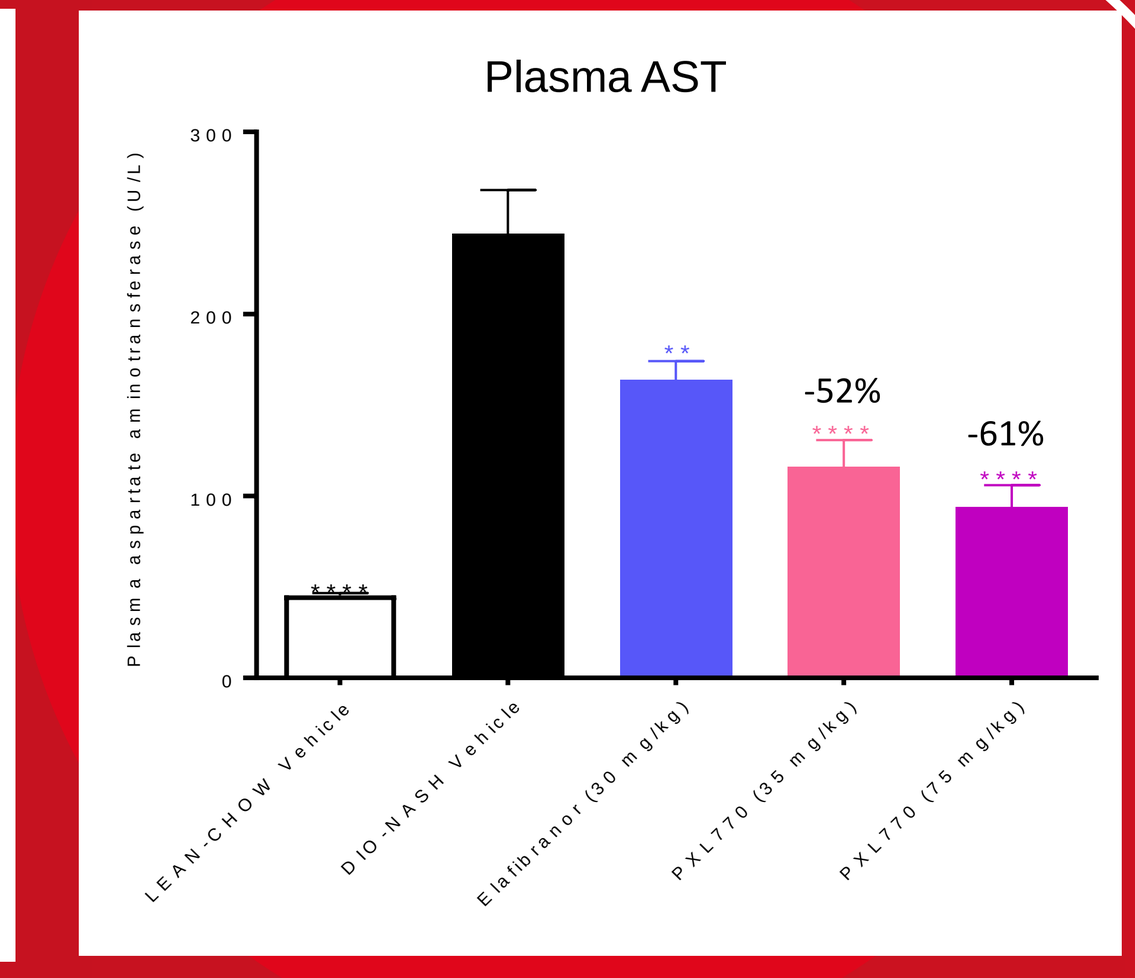
Plasma AST
Plasma aspartate aminotransferase (U/L)
-52%
-61%
LEAN-CHOW Vehicle
DIO-NASH Vehicle
Elafibranor (30 mg/kg)
PXL770 (35 mg/kg)
PXL770 (75 mg/kg)

*p<0.05, **P<0.01, ***P<0.001, ****P<0.0001 vs. DIO-NASH Vehicle

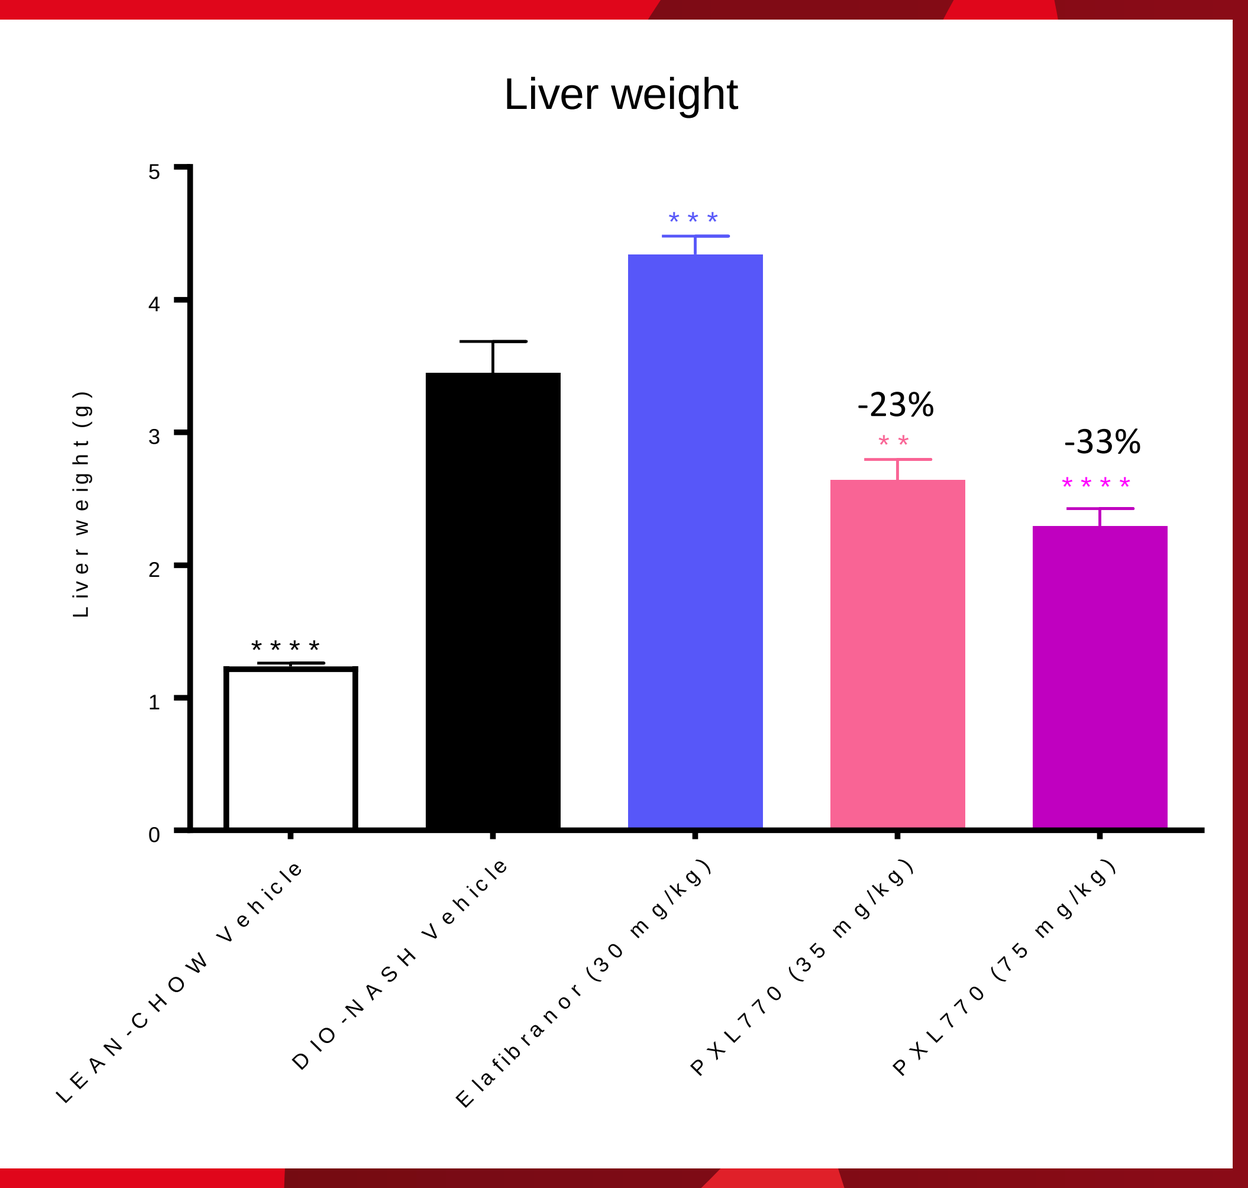
Liver weight
Liver weight (g)
-23%
-33%
LEAN-CHOW Vehicle
DIO-NASH Vehicle
Elafibranor (30 mg/kg)
PXL770 (35 mg/kg)
PXL770 (75 mg/kg)

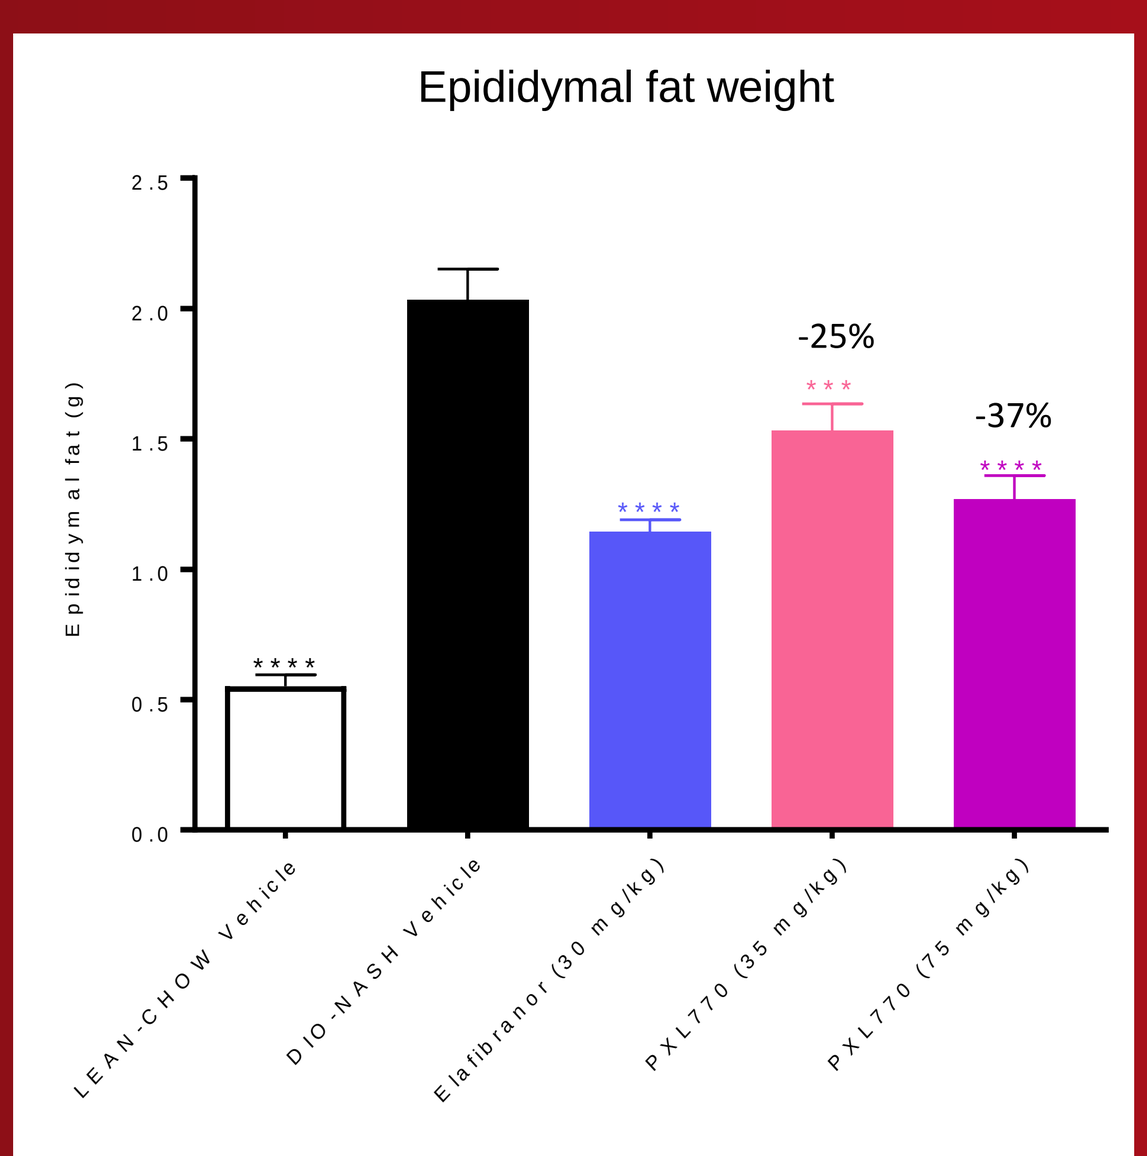
Epididymal fat weight
Epididymal fat (g)
-25%
-37%
LEAN-CHOW Vehicle
DIO-NASH Vehicle
Elafibranor (30 mg/kg)
PXL770 (35 mg/kg)
PXL770 (75 mg/kg)

Figure 2 - DIO-NASH mice demonstrated significant increase in body weight (38g vs. 29g in Lean mice, p<0.001), liver weight and epididymal fat depot weight, increase in levels of plasma cholesterol and free fatty acids (FFA) and levels of liver enzymes. PXL770 slightly reduced body weight at the highest dose (-5%) and reduced liver and epididymal fat depot weights at both doses. PXL770 decreased significantly plasma cholesterol and FFA as well as liver enzymes, ALT and AST

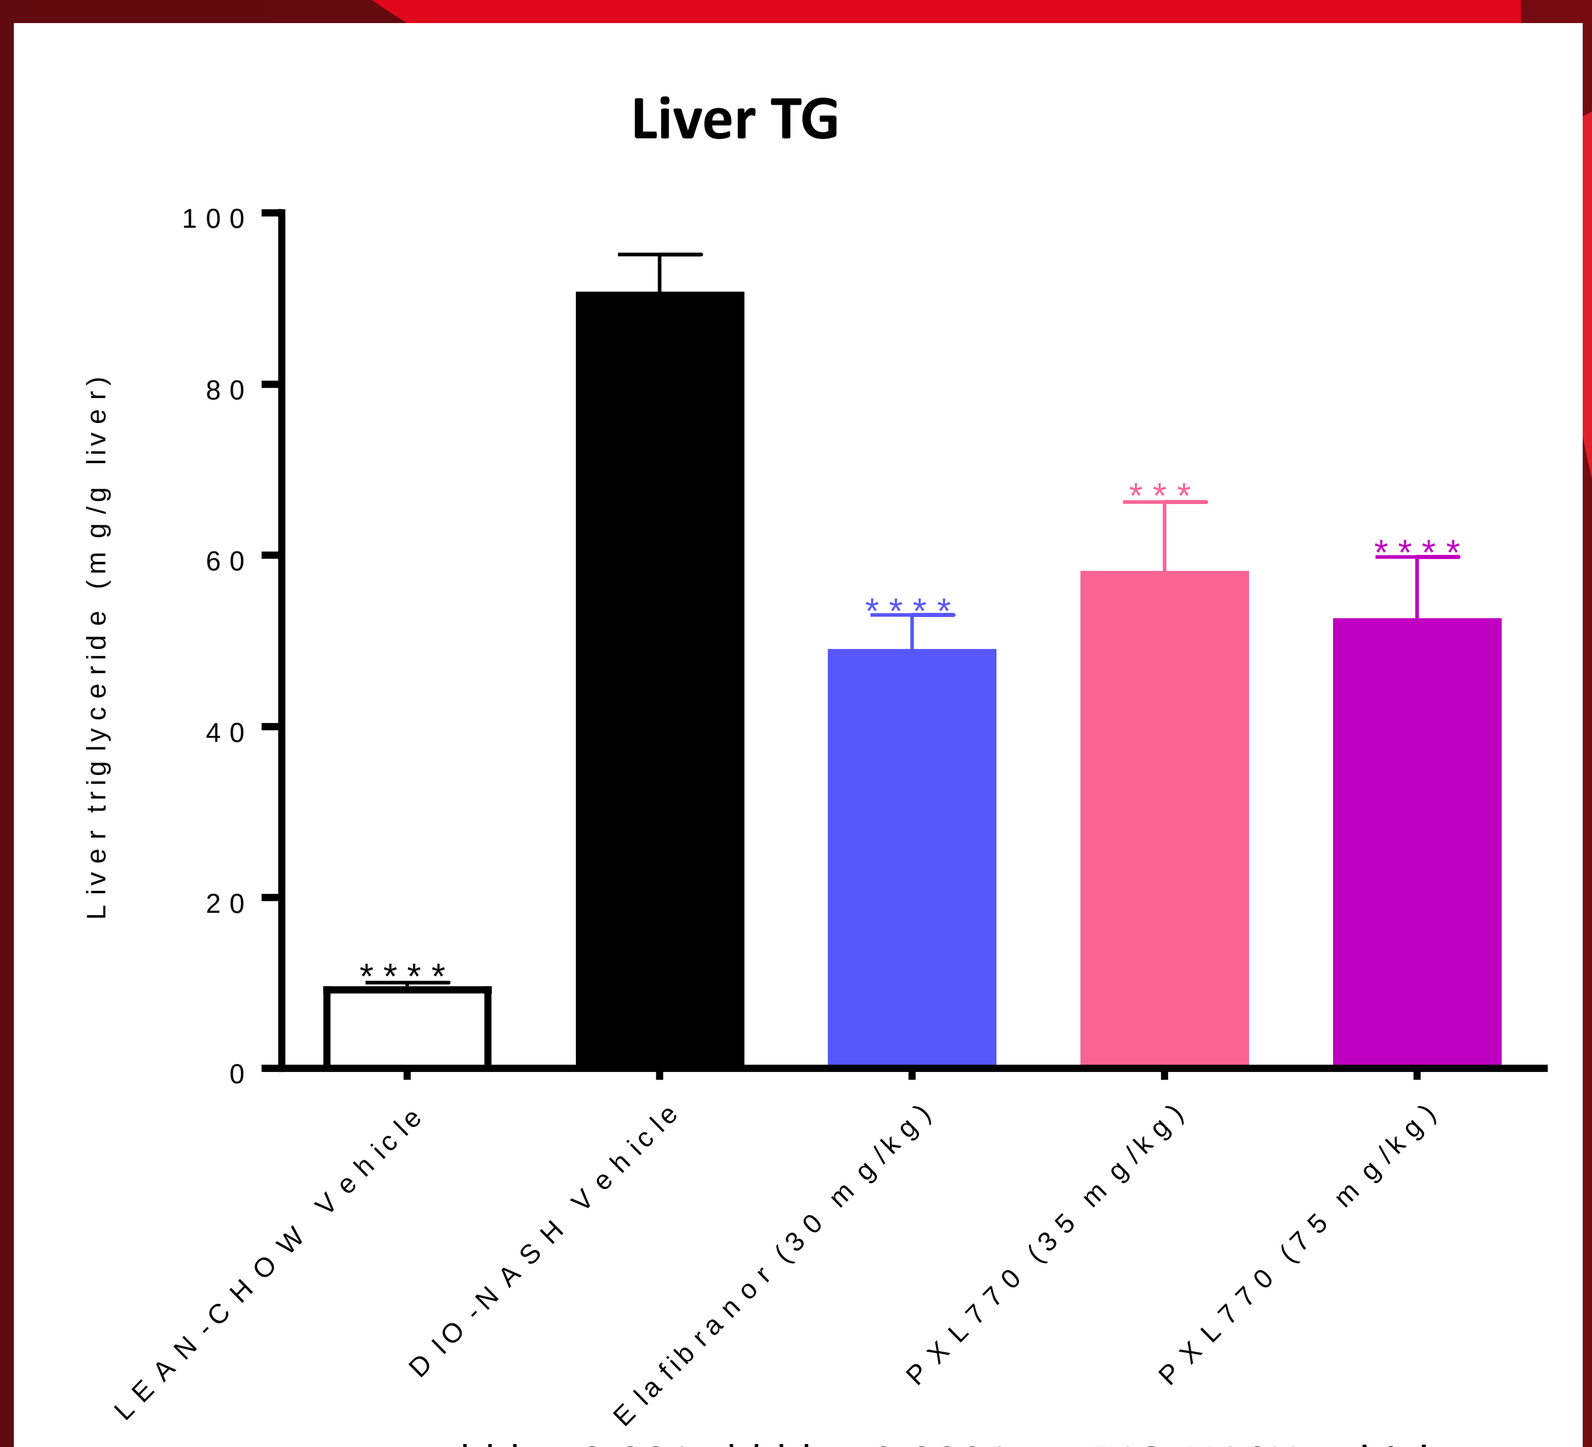
Liver TG
Liver triglyceride (mg/g liver)
LEAN-CHOW Vehicle
DIO-NASH Vehicle
Elafibranor (30 mg/kg)
PXL770 (35 mg/kg)
PXL770 (75 mg/kg)

p<0.001, *p<0.0001 vs. DIO-NASH vehicle

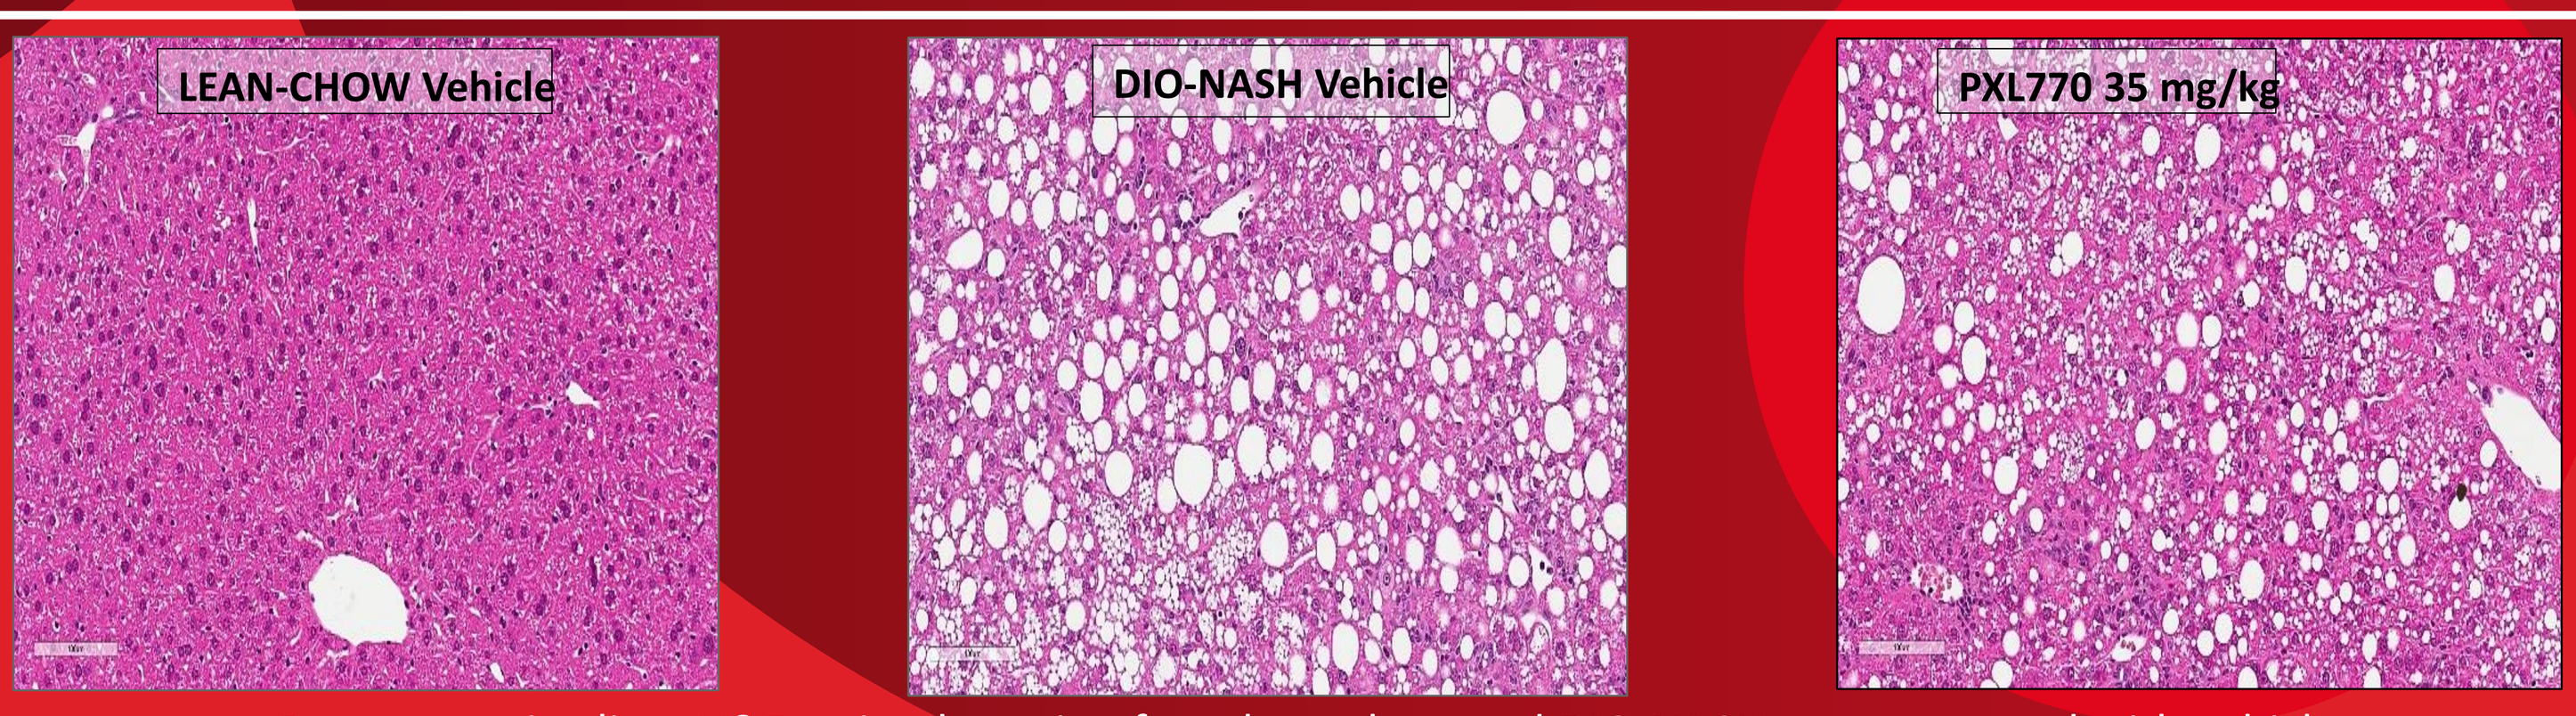
LEAN-CHOW Vehicle
DIO-NASH Vehicle
PXL770 35 mg/kg

Representative liver H&E stained section from lean-chow and DIO-NASH mouse treated with vehicle and DIO-NASH mouse treated with elafibranor (30 mg/kg/d) or PXL770 (35 or 75 mg/kg twice daily)

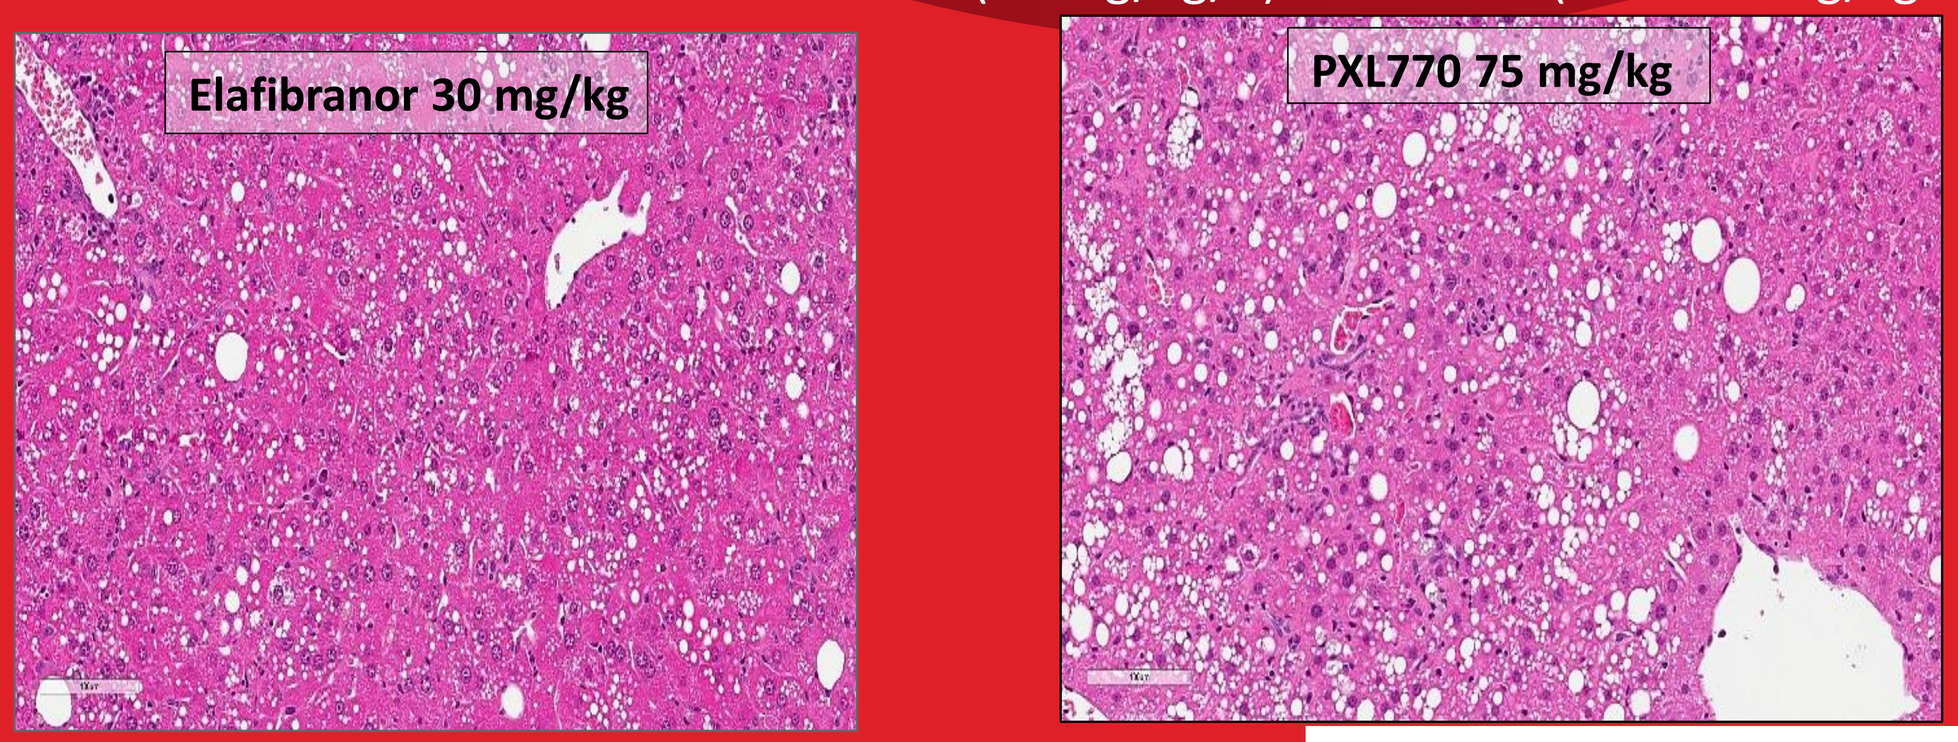
Elafibranor 30 mg/kg
PXL770 75 mg/kg

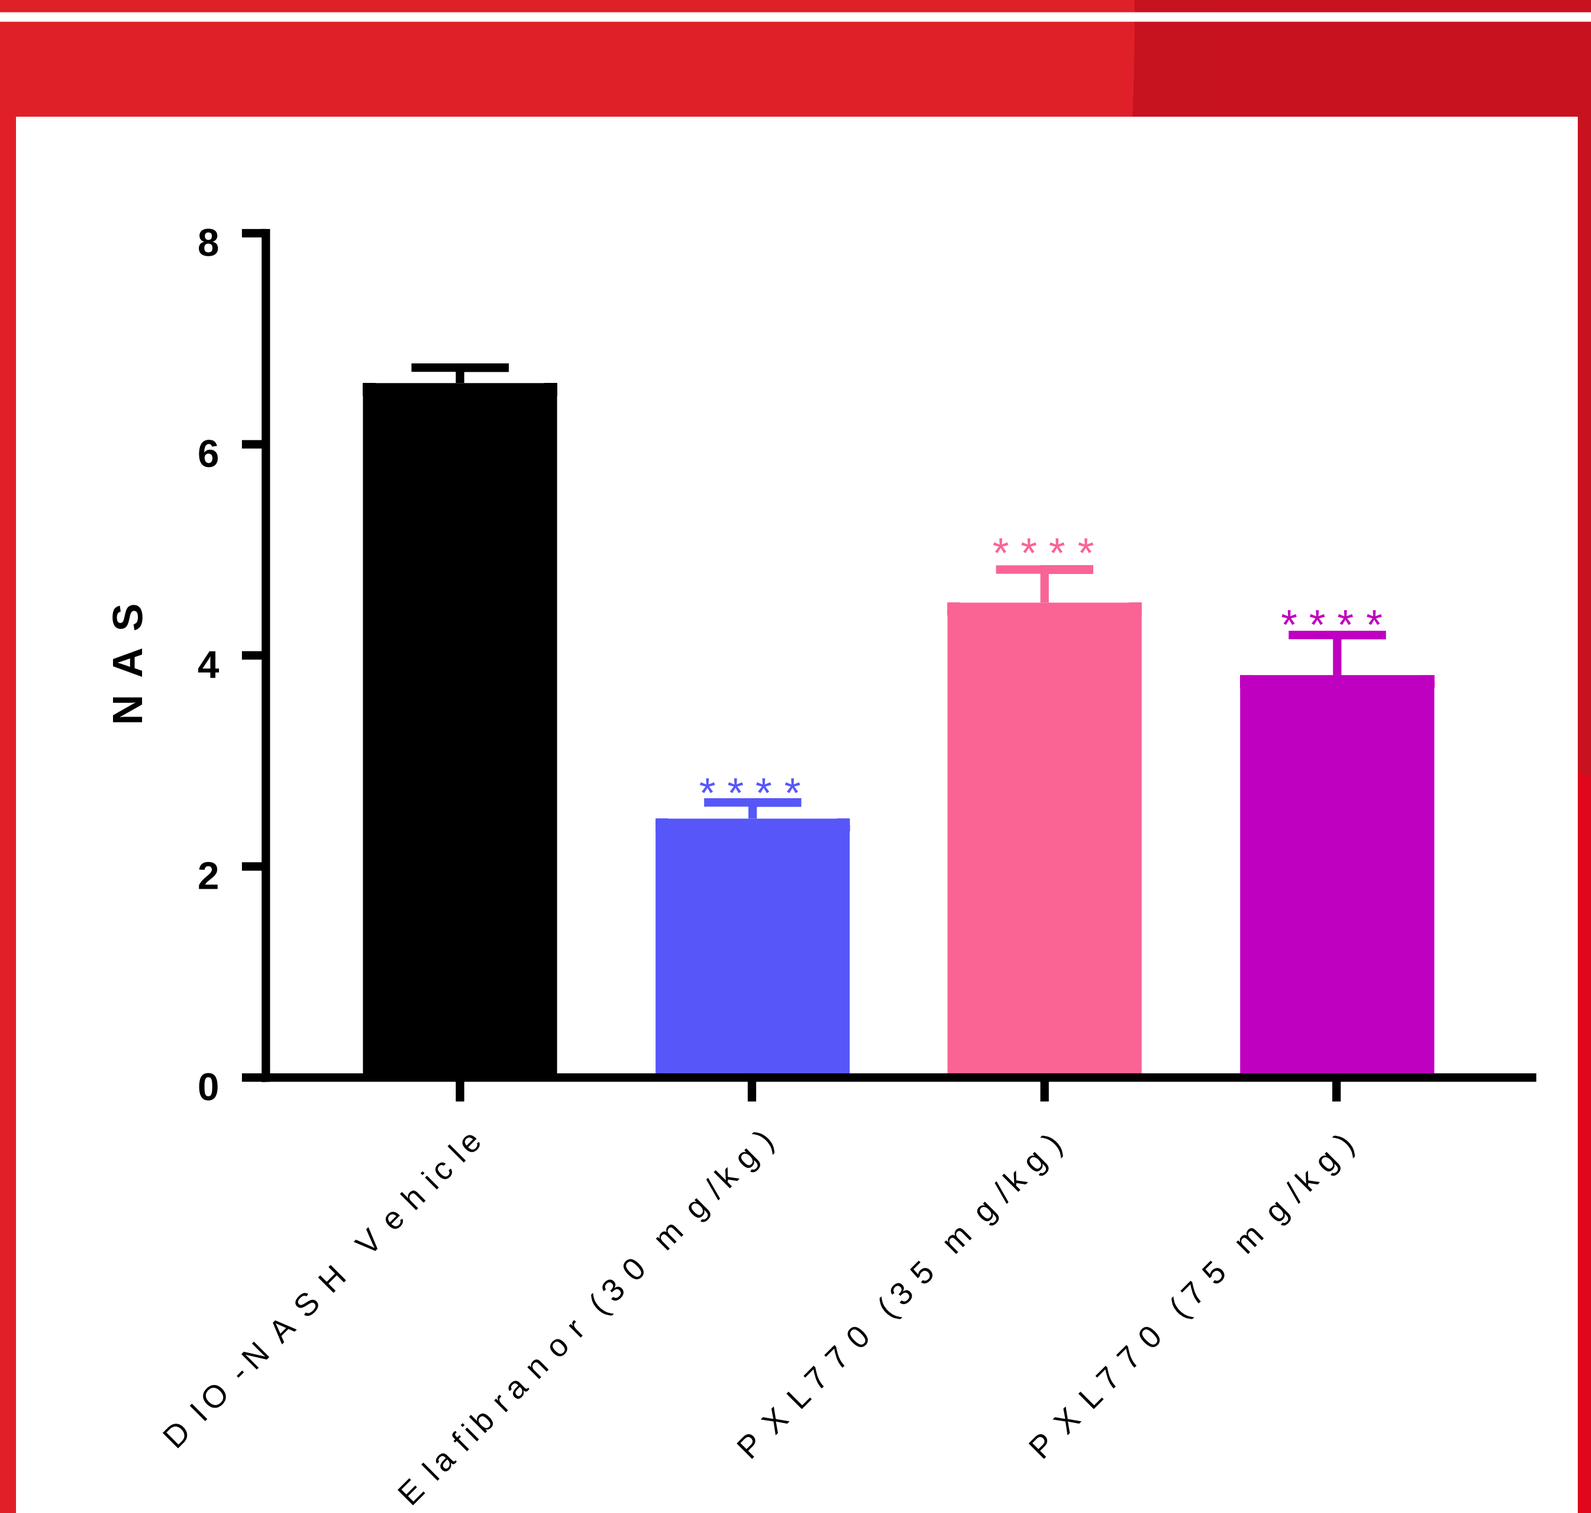
NAS
DIO-NASH Vehicle
Elafibranor (30 mg/kg)
PXL770 (35 mg/kg)
PXL770 (75 mg/kg)

****p<0.0001 vs. DIO-NASH vehicle

Figure 3 – DIO-NASH mice demonstrated significant elevated NAS (H&E staining) accompanied by an increase in liver triglycerides.

Both PXL770 doses reduced significantly NAS (-36% and -46% p<0.001 at 35 and 75 mg/kg respectively) decreasing Steatosis, Inflammation and hepatic Ballooning.

The benefit on liver steatosis was confirmed by the reduction of liver triglycerides (-36% and -42% p<0.001 at 35 and 75 mg/kg respectively)

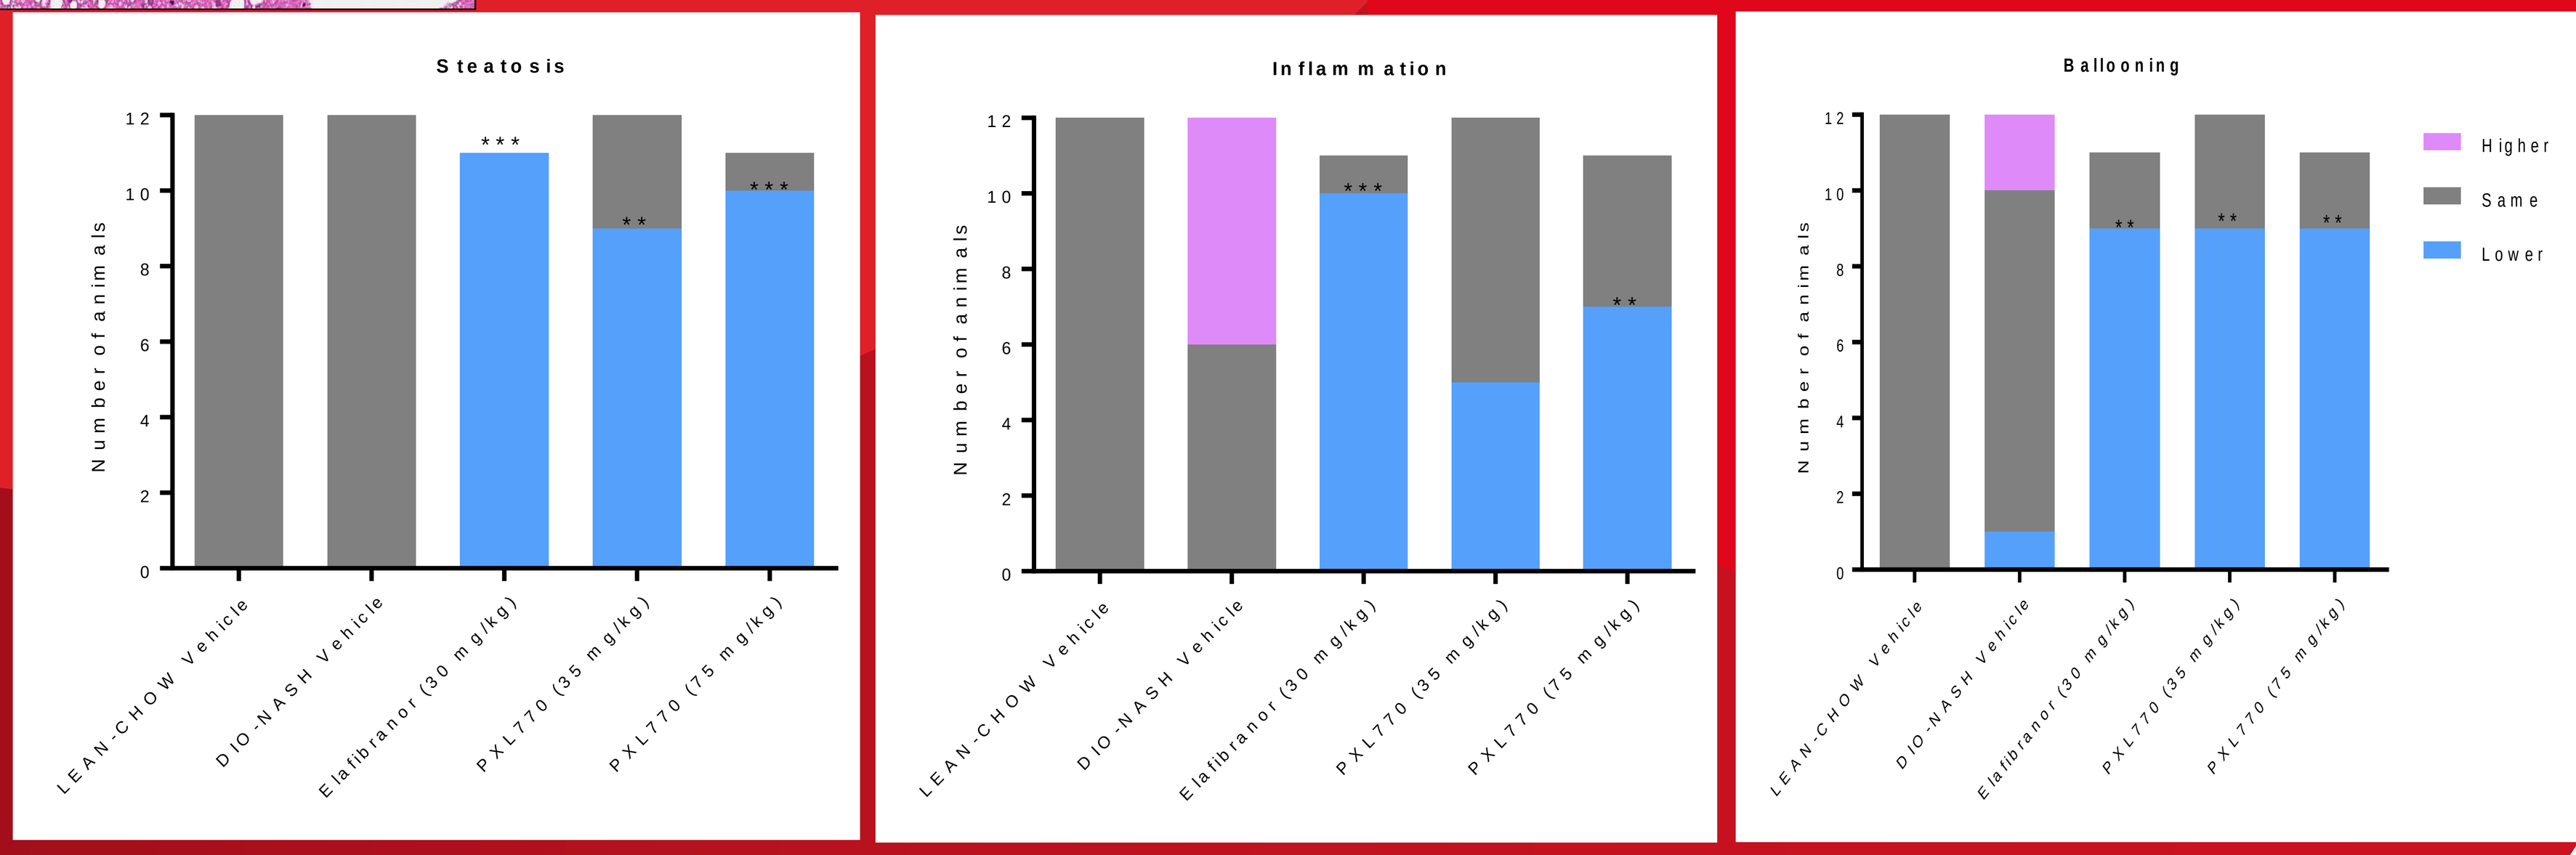
Steatosis
Inflammation
Ballooning
Number of animals
Higher
Same
Lower
LEAN-CHOW Vehicle
DIO-NASH Vehicle
Elafibranor (30 mg/kg)
PXL770 (35 mg/kg)
PXL770 (75 mg/kg)

The number of animals with a higher (worsening), same or lower (improvement) in score at post- compared to pre-study is indicated by the height of the bar. **p<0.01, ***p<0.001 vs. DIO-NASH vehicle

Figure 4 – DIO-NASH mice demonstrated significant elevated fibrosis score. PXL770 strongly down-regulated a panel of genes expression involved in fibrosis e.g. type I (-68%) and type III (-63%) collagen genes expression, suggesting that a longer term treatment could lead to a benefit of PXL770 on fibrosis

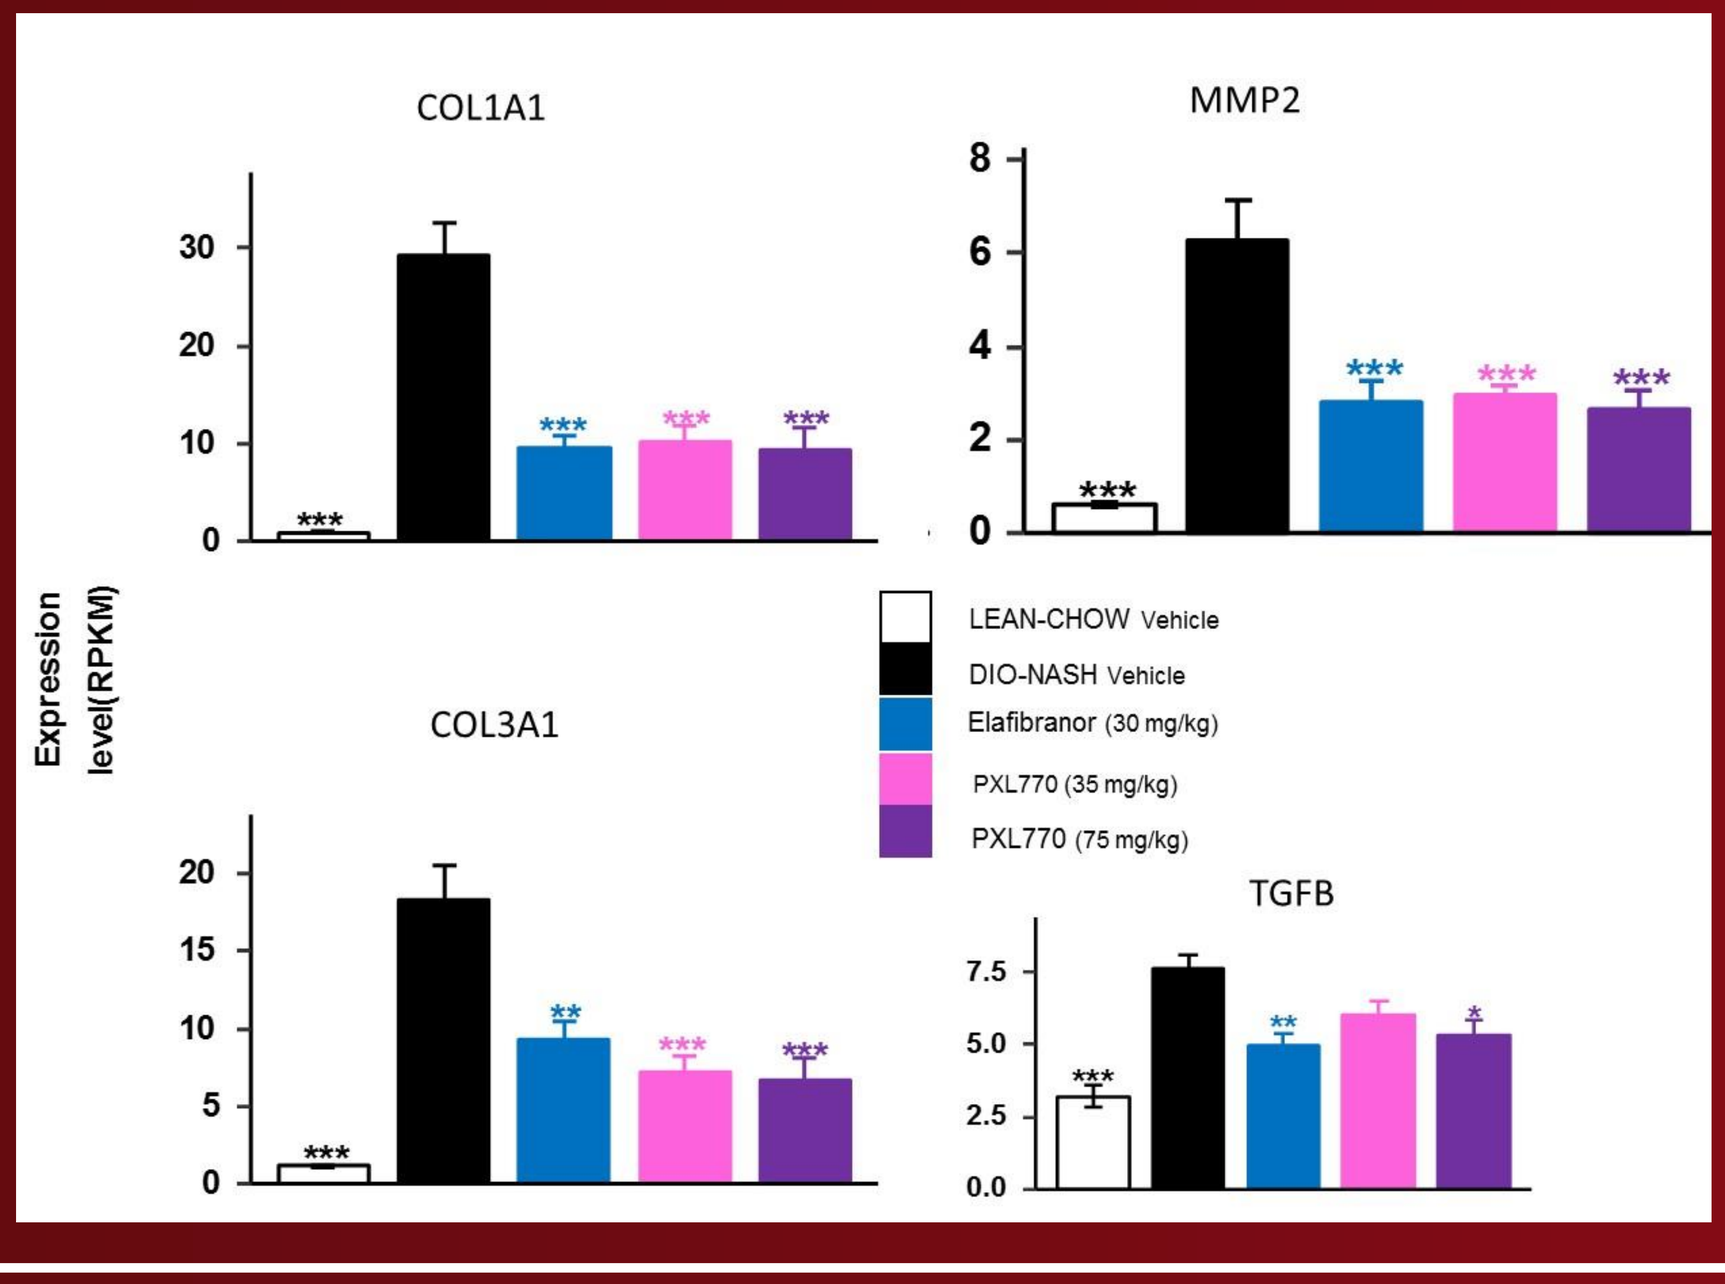
COL1A1
MMP2
COL3A1
TGFB
Expression level(RPKM)
LEAN-CHOW Vehicle
DIO-NASH Vehicle
Elafibranor (30 mg/kg)
PXL770 (35 mg/kg)
PXL770 (75 mg/kg)

p<0.01, *p<0.001, ****p<0.0001 vs. DIO-NASH vehicle

Conclusion

In this DIO-NASH mouse model, PXL770 by directly activating AMPK:

- Improved metabolic parameters (plasma cholesterol, FFA, liver enzymes)
- Reduced liver weight and epididymal fat depot weight
- Reduced NAFLD activity score and liver triglycerides content
- Reduced fibrotic genes expression

According to these results, PXL770 appears promising for the treatment of NAFLD/NASH

GLOBAL NASH CONGRESS 2018 – LONDON 26-27 FEBRUARY 2018

poxel
Excellence in achieving PoC