Safety and Efficacy of Preoperative Meloxicam IV in Colorectal Surgery

PRESENTER: **Stewart McCallum, MD**

BACKGROUND: Intravenous (IV) meloxicam (Meloxicam IV; Anjeso™) is approved for use in adults for the management of moderate-to-severe pain, alone or in combination with non-NSAID analgesics. Limitation of Use - Because of delayed onset of analgesia, meloxicam IV alone is not recommended for use when rapid onset of analgesia is required. Clinical studies supporting approval were conducted in the postoperative setting. This study involves dosing preoperatively in subjects undergoing colorectal procedures.

METHODS

- Phase 3b, placebo controlled (1:1) study in subjects undergoing bowel resection and/or anastomosis.
- Subjects randomized to meloxicam IV 30 mg or placebo with first dose 30 minutes before start of surgery, then every 24 hours on top of a Standardized Enhanced Recovery After Surgery (ERAS) protocol.
 - Preoperative hydration, gabapentin, acetaminophen.
 - Alvimopan prohibited
- Opioid rescue available on request

RESULTS

Table 1: Summary of Adverse Events

Category / Term	Meloxicam IV 30 mg N=27 n (%)	Placebo N=28 n (%)
Any TEAE	23 (85.2)	26 (92.9)
Any SAE	3 (11.1)	4 (14.3)
Withdrawn due to AE	0	0
Deaths	0	0
Most Common AEs		
Nausea	9 (33.3)	14 (50.0)
Vomiting	5 (18.5)	5 (17.9)
Hypokalemia	2 (7.4)	7 (25.0)
Hypophosphatemia	2 (7.4)	6 (21.4)
Ileus	1 (3.7)	5 (17.9)
Hypertension	2 (7.4)	3 (10.7)

TEAE=Treatment-Emergent Adverse Event; SAE = Serious Adverse Event; AE=Adverse Event

Safety assessments comparable between meloxicam IV 30 mg once daily and placebo.[Table1]

Inpatient **<u>opioid use reduced by 35.3%</u>** in meloxicam IV group.[Fig 1]

Significant reduction in time to:

<u>first bowel sounds</u> (↓58.9%)[Fig 2],

<u>first bowel movement</u> (↓17.5%)[Fig 2], and

<u>hospital discharge</u> (↓27.0%)[Fig 3]

for meloxicam IV vs. placebo group (p<0.05)

MED.0320-136.a

Figure 1: Opioid Consumption: End of Surgery through Discharge

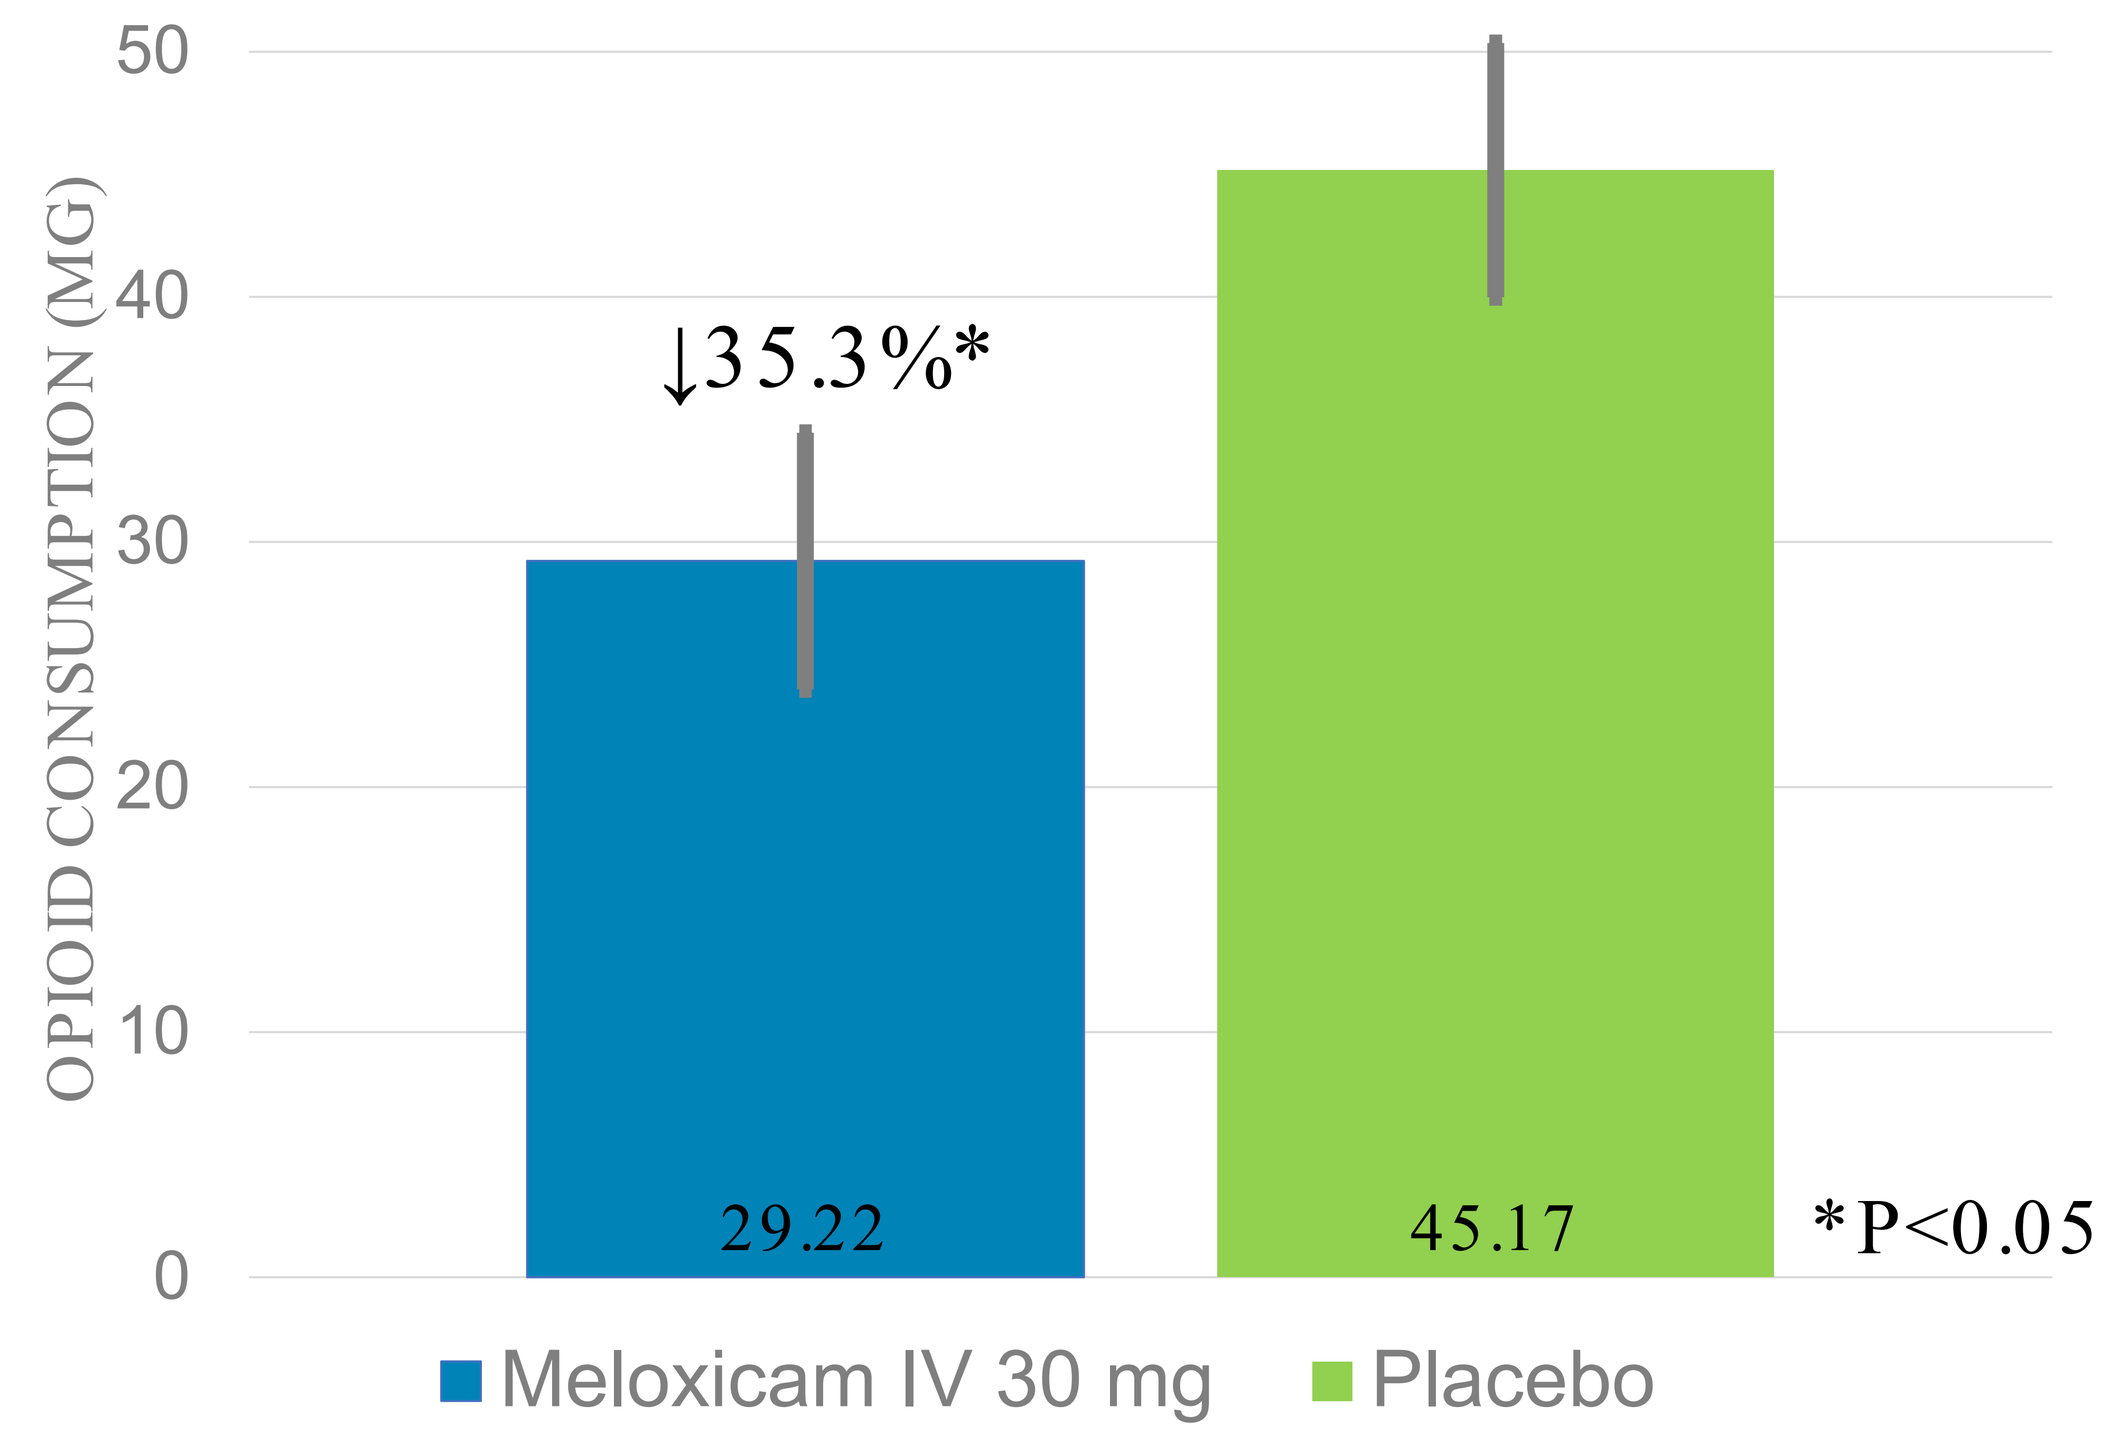
OPIOID CONSUMPTION (MG)
50
40
30
20
10
0
↓35.3%*
29.22
45.17
*P<0.05
Meloxicam IV 30 mg
Placebo

Figure 2: Time to Bowel Function Recovery

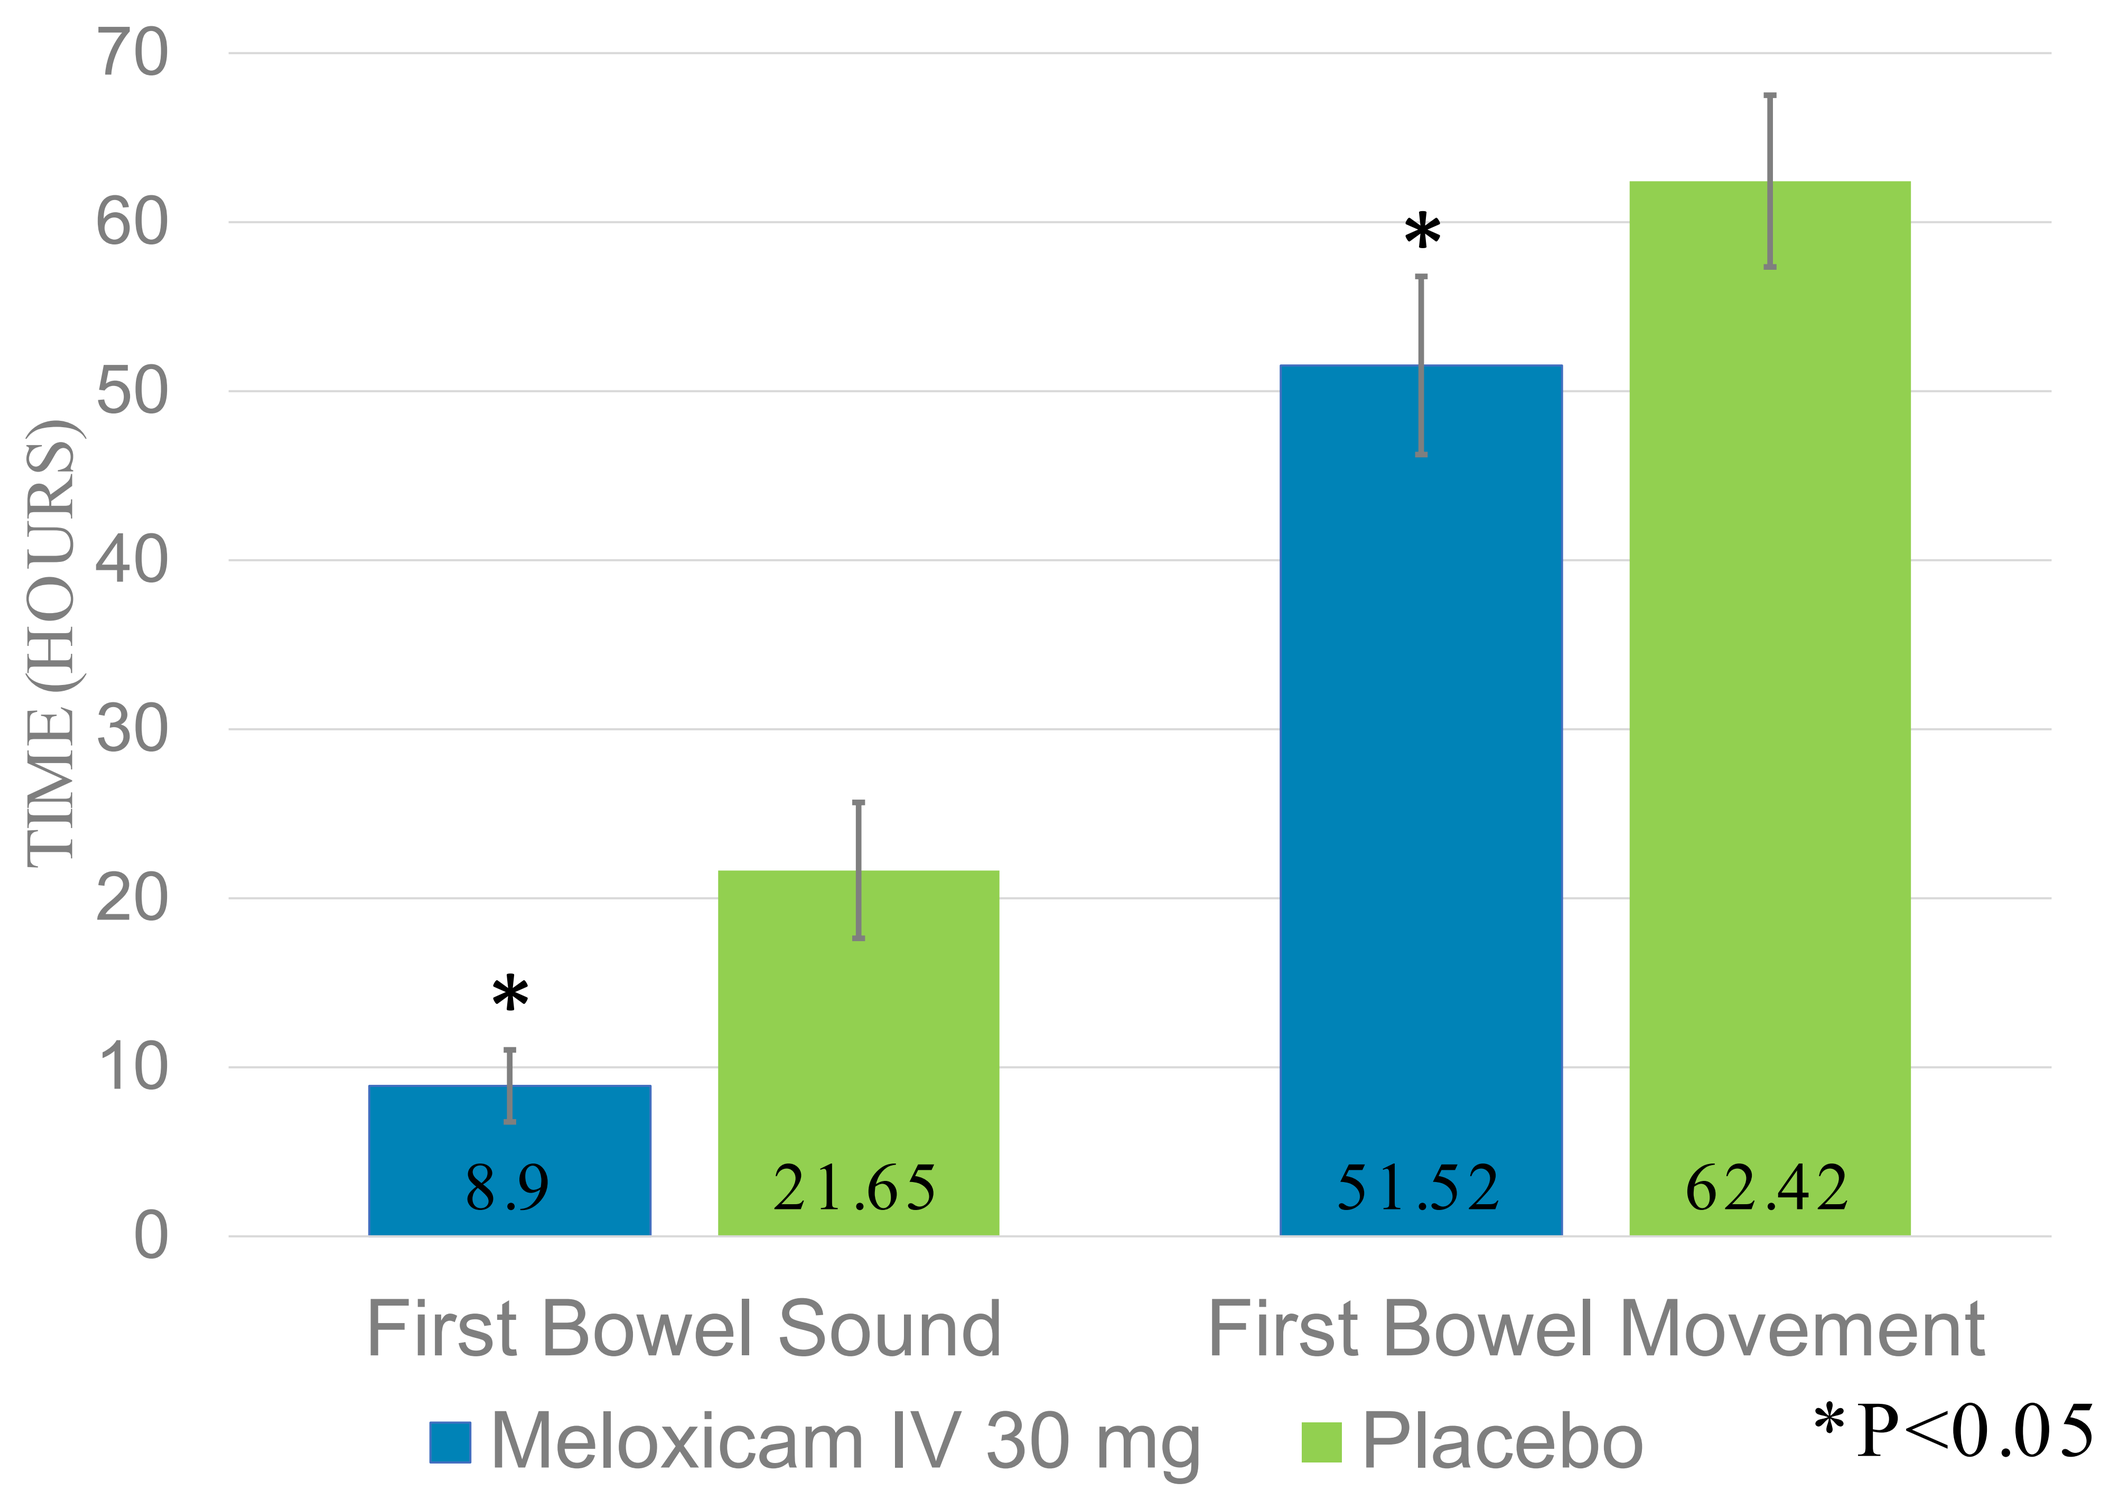
TIME (HOURS)
70
60
50
40
30
20
10
0
*
*
8.9
21.65
51.52
62.42
First Bowel Sound
First Bowel Movement
Meloxicam IV 30 mg
Placebo
*P<0.05

Figure 3: Time toHospital Discharge Order

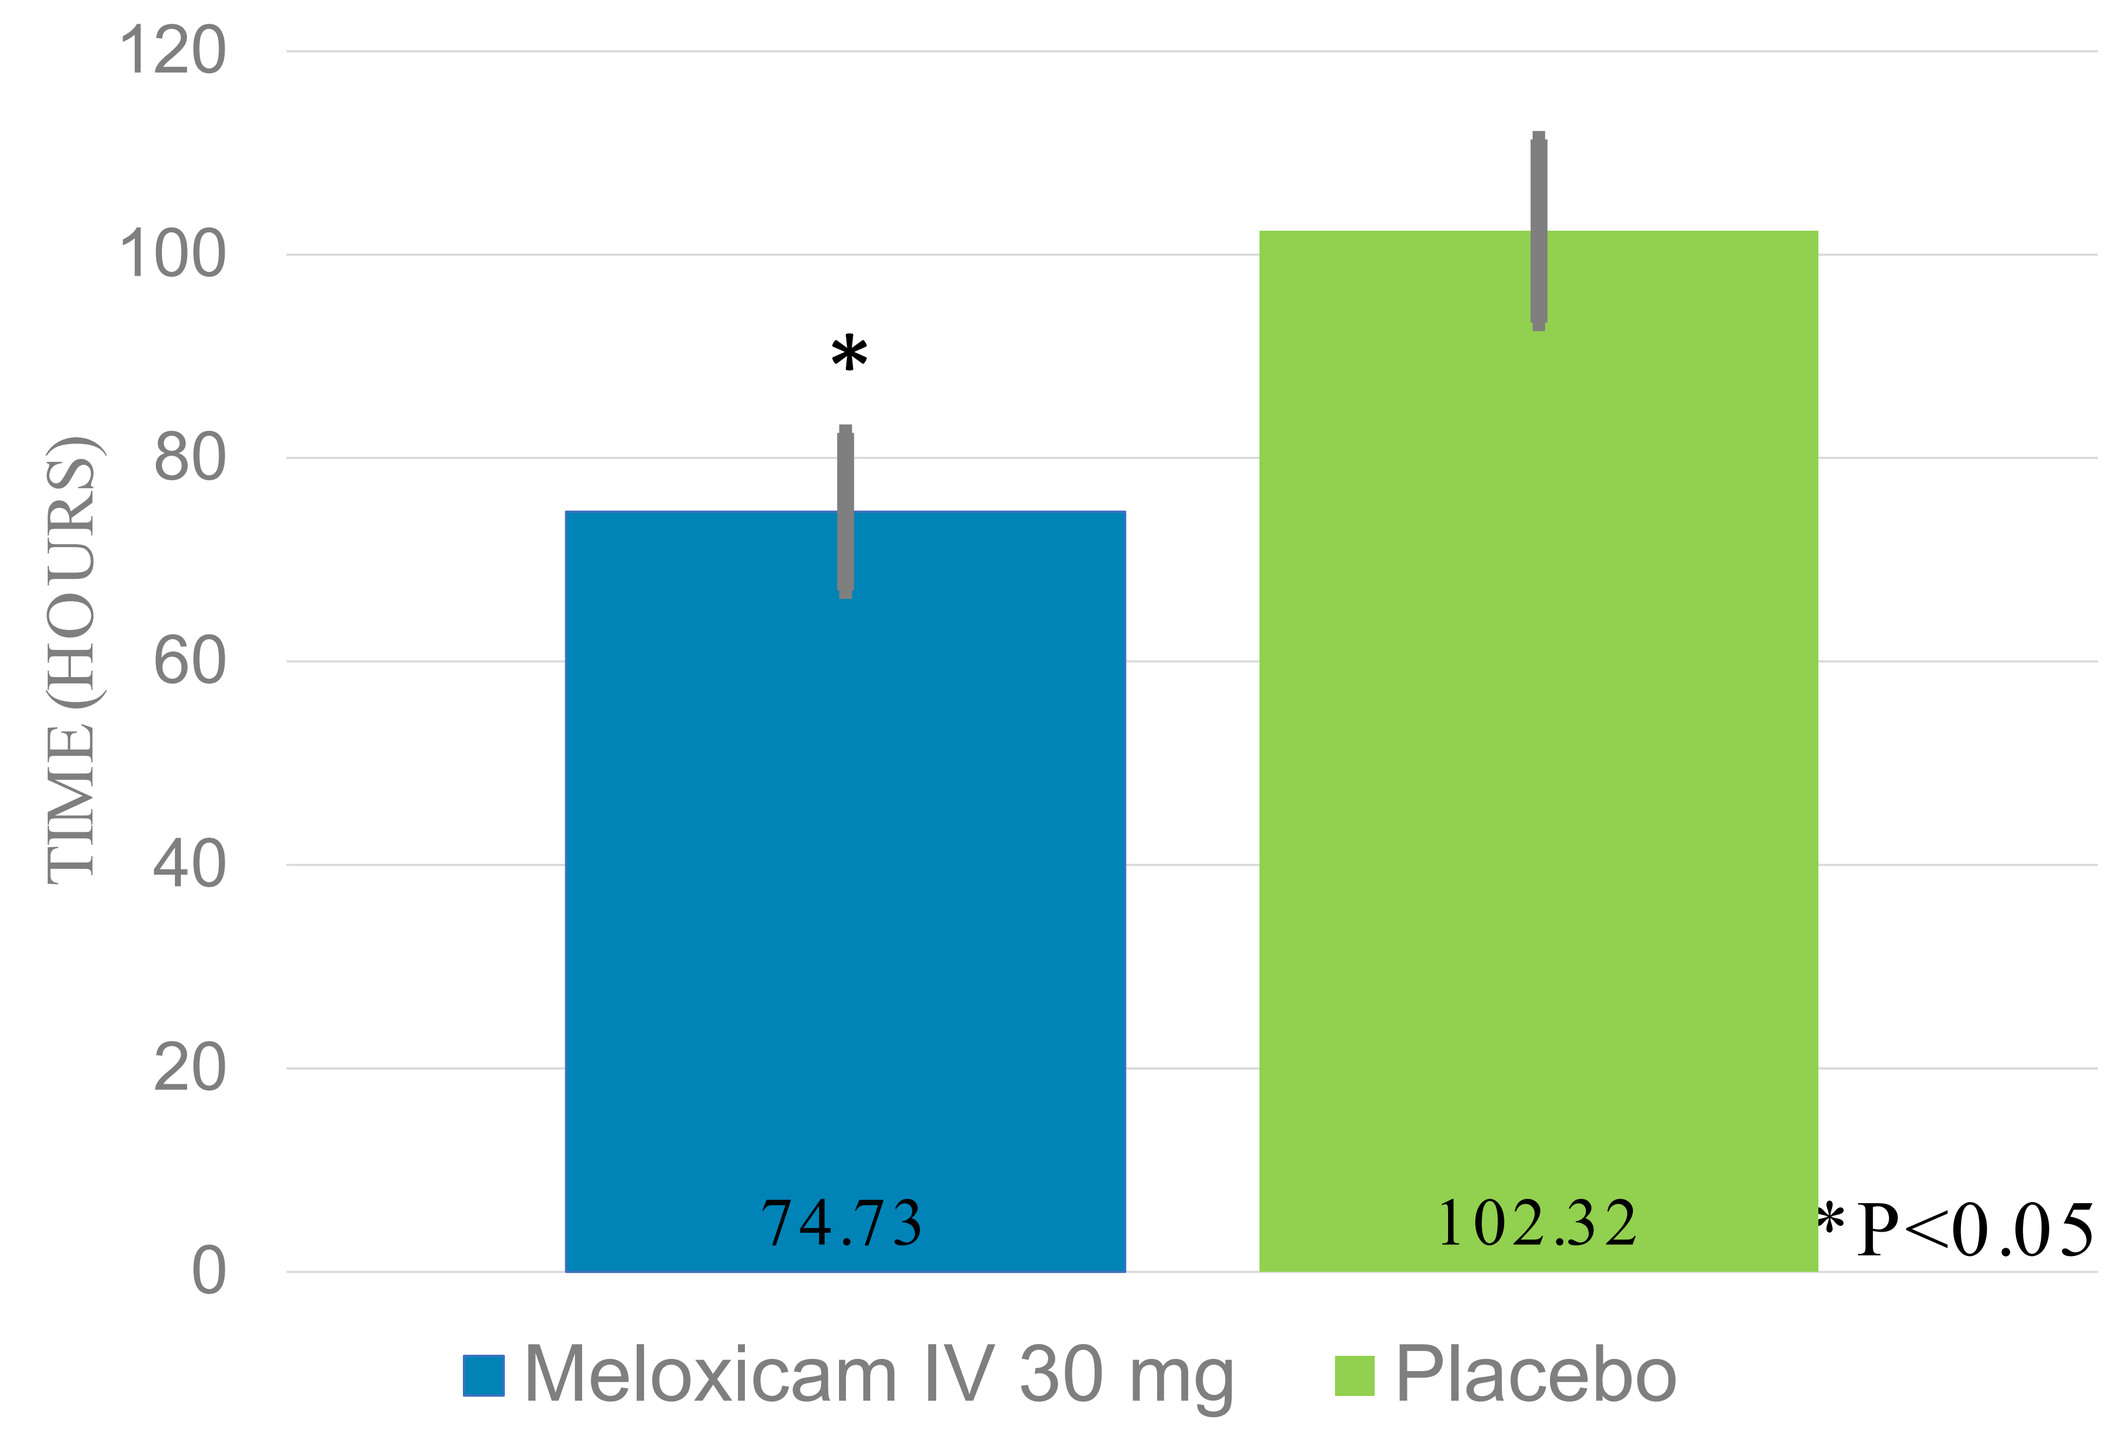
TIME (HOURS)
120
100
80
60
40
20
0
*
74.73
102.32
*P<0.05
Meloxicam IV 30 mg
Placebo

Conor Delaney[1], Jennifer Silinsky[2], Stewart McCallum[3], Randall Mack[3], Alex Freyer[3], Teresa Galas[3], Libby Black[3], Wei Du[4]

[1] Dept of Colorectal Surgery, Cleveland Clinic, Cleveland, OH
[2] Dept of Surgery, Tulane University, New Orleans, LA
[3] Baudax Bio, Inc., Malvern, PA
[4] Clinical Statistics Consulting, Blue Bell, PA

E-poster P1047