Novel Induced-Mesenchymal Stem Cells (*i*-MSCs) Attenuate Severity of ARDS in Septic Sheep

peenkhba@utmb.edu

K. Hashimoto[1], N. Bazhanov[1], P. Enkhbaatar[1], M. Angel[2], A. Lader[3], M. Czuczman[4], and M. Matthay, [5]

[1] University of Texas Medical Branch Galveston, TX; [2] Novellus Therapeutics, Cambridge, MA; [3] Citius Pharma, Cranford, NJ; [4] NoveCite, Inc, Cranford, NJ; [5] UCSF Department of Medicine and Department of Anesthesia, Cardiovascular Research Institute, San Francisco, CA

Abstract or Introduction

Acute respiratory distress syndrome (ARDS) is respiratory failure resulting from noncardiogenic pulmonary edema. ARDS is commonly caused by bacterial or viral pneumonia. ARDS is associated with very high mortality (36-45%) (1).

Currently there is no FDA approved drug therapy for ARDS. A growing body of evidence supports mesenchymal stem cell (MSC)-based therapy as a potential treatment for ARDS. MSCs target a large number of dysregulated inflammatory cytokines and provide tissue repair and pathogen clearing capabilities via multimodal mechanisms-of-action. Previously, we have demonstrated beneficial effects of bone marrow (BM)-derived MSCs in an ovine model of lung injury (2).

Although BM-MSCs have shown excellent clinical safety, they have struggled to demonstrate therapeutic efficacy in many diseases of interest.

High efficiency and lack of vector persistence make mRNA reprogramming an ideal platform for the development of new autologous and allogeneic MSC therapies. Previously, we have shown that *i*-MSCs exhibit superior growth potential, secretome, and therapeutic efficacy in a demyelinating disease model compared to BM-MSCs (3), suggesting that *i*-MSCs may be an ideal cell therapy for devastating inflammatory diseases, such as sepsis induced ARDS.

Objectives

Objective: To test the safety and efficacy of a novel potent iPSC-induced MSCs (*i*-MSCs) in a clinically relevant sheep model of sepsis-induced ARDS.

Hypothesis: *i*-MSCs reduce the severity of lung injury and will be well tolerated in sheep model of ARDS induced by instillation of *Ps. aeruginosa* into lungs.

Methods

Preparation of induced pluripotent stem cell-derived MSCs (*i*-MSCs):

First, iPSCs were generated from adult human fibroblasts derived from a dermal punch biopsy through a one-week, high efficiency, immunosuppressant-free reprogramming protocol using mRNA encoding Oct4, Sox2, Klf4, c-Myc, and Lin28 (A). These cells expressed pluripotency factors (B).

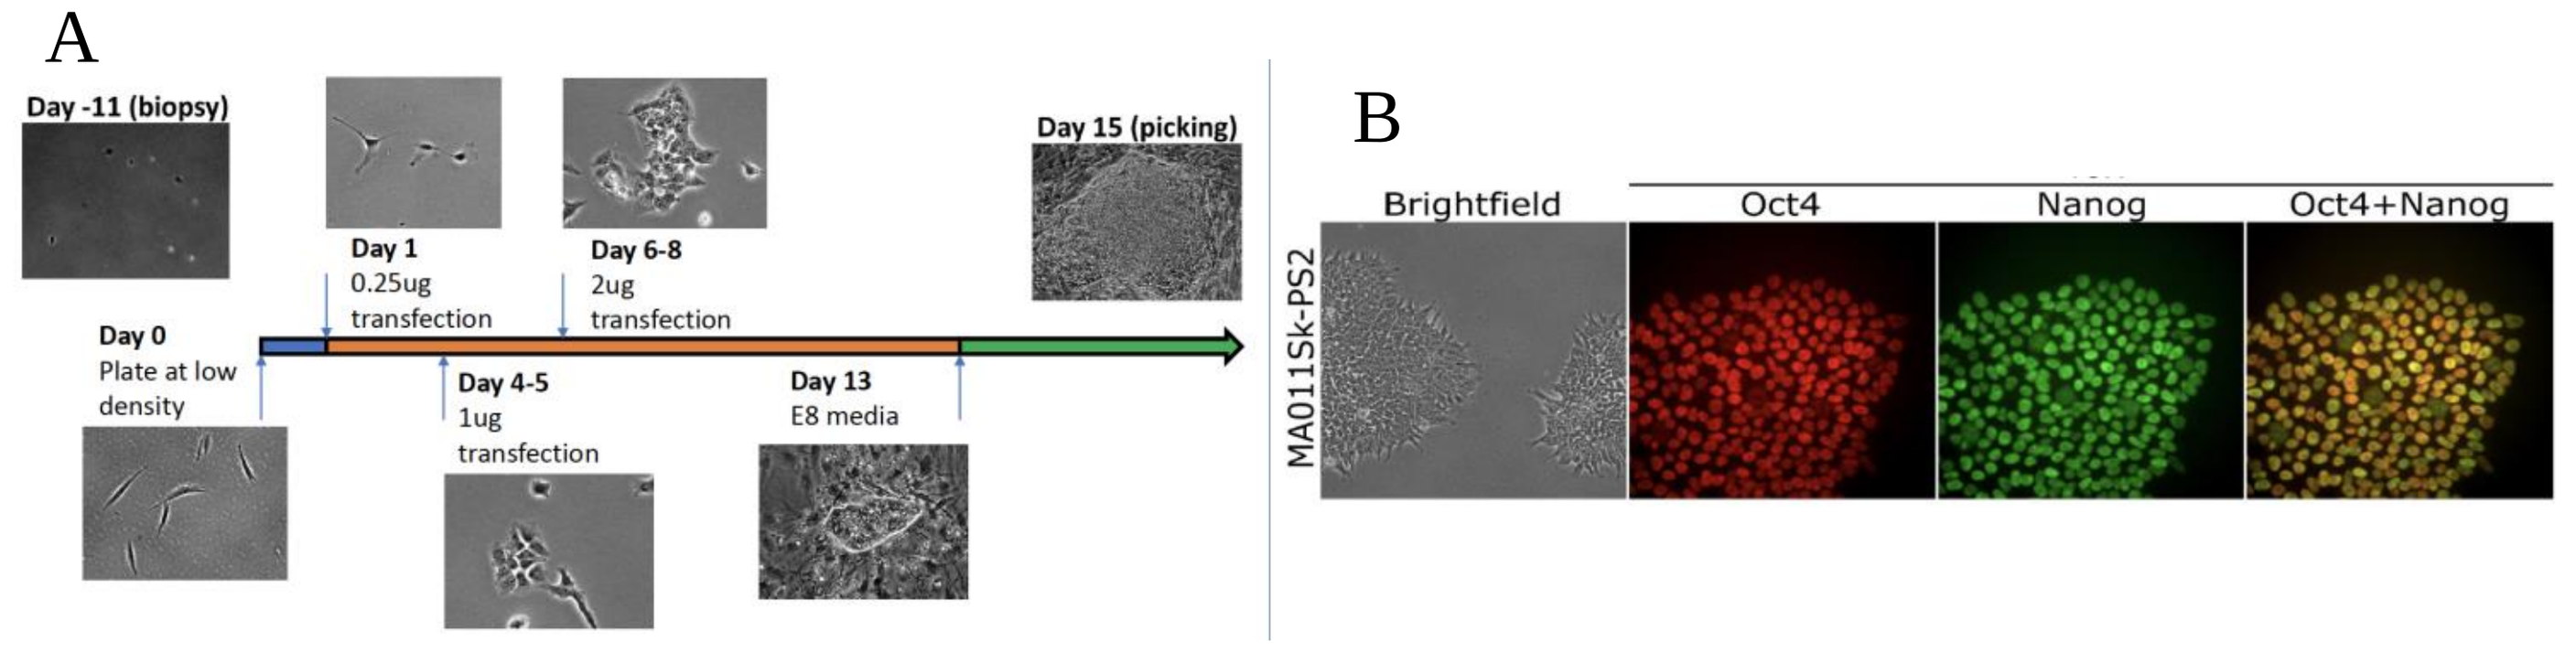

A
Day -11 (biopsy)
Day 0
Plate at low density
Day 1
0.25ug transfection
Day 4-5
1ug transfection
Day 6-8
2ug transfection
Day 13
E8 media
Day 15 (picking)
B
Brightfield
Oct4
Nanog
Oct4+Nanog
MA011Sk-PS2

Second, *i*-MSCs were generated using the STEMdiff Mesenchymal Progenitor Kit (STEMCELL Technologies) (A). *i*-MSCs were differentiated to adipocytes, osteoblasts and chondrocytes (B). *i*-MSCs underwent 74.8 population doublings before senescence compared to 18.2 population doublings for the BM-MSCs (C).

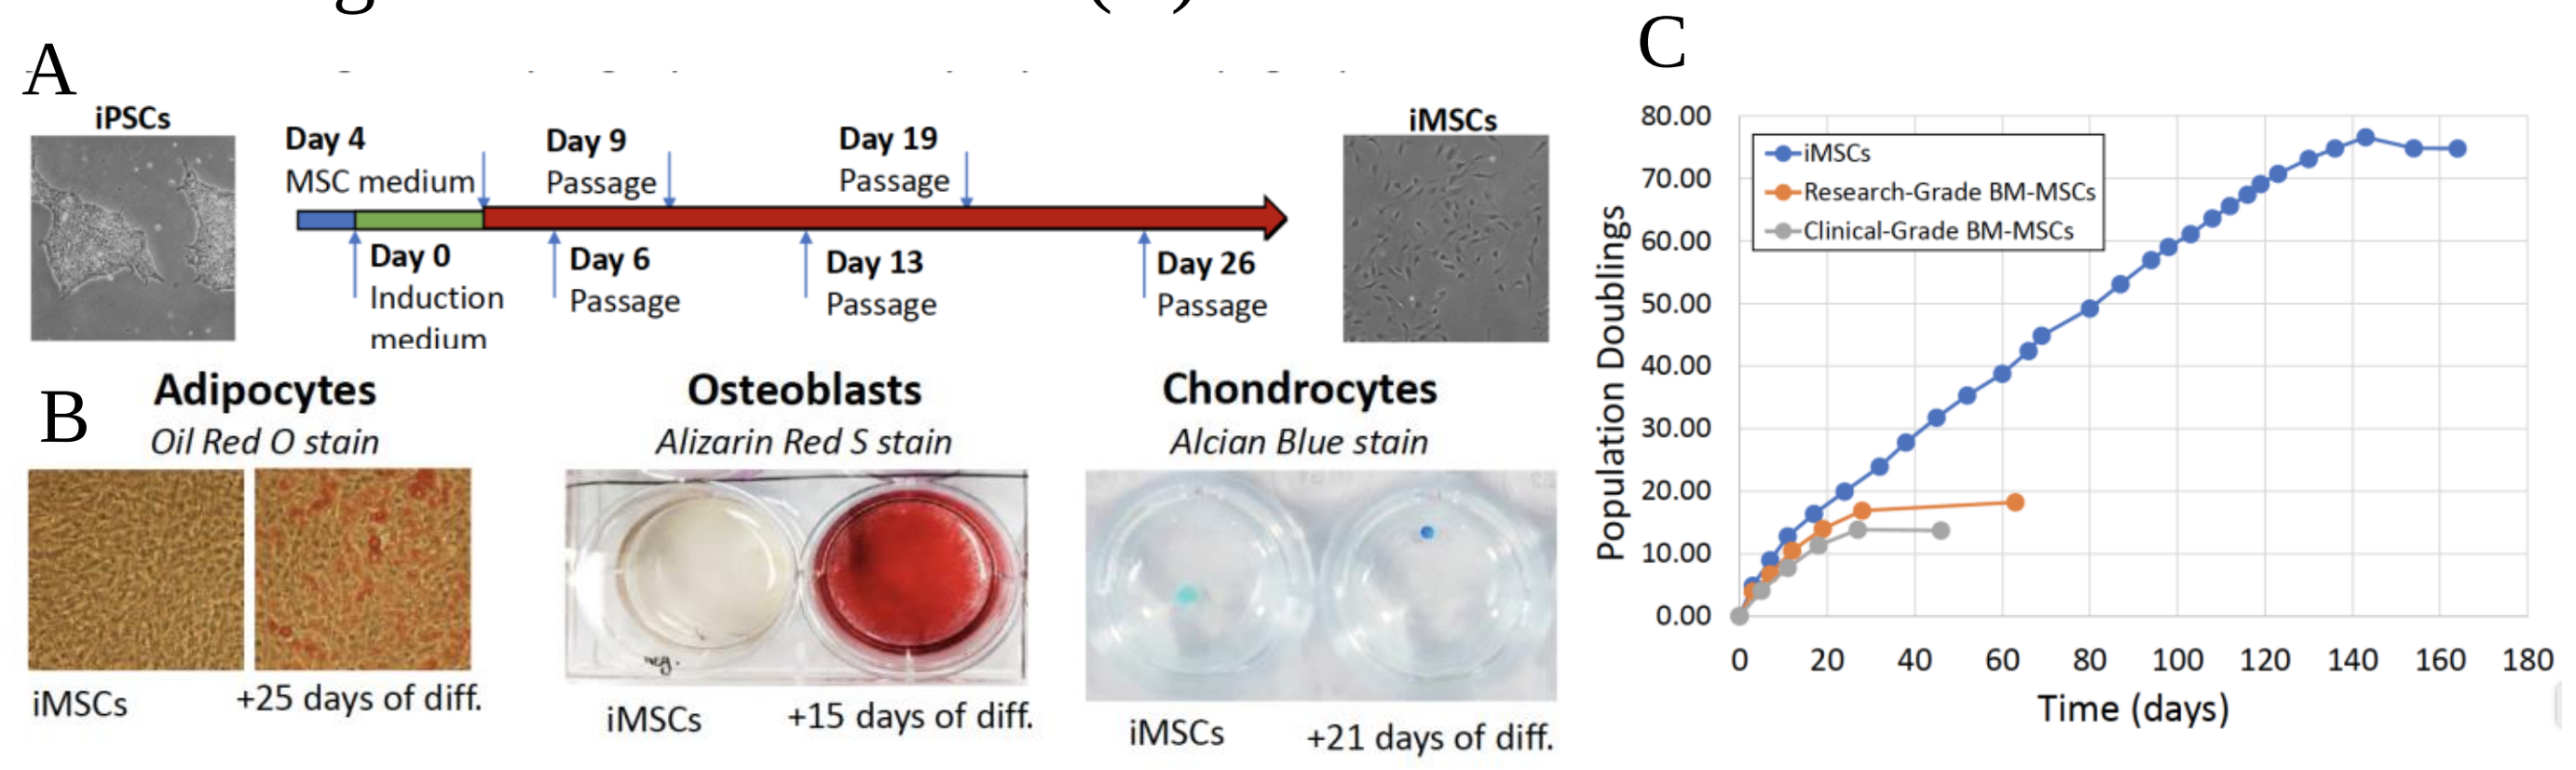

A
iPSCs
Day 4
MSC medium
Day 0
Induction medium
Day 6
Passage
Day 9
Passage
Day 13
Passage
Day 19
Passage
Day 26
Passage
iMSCs
B
Adipocytes
Oil Red O stain
Osteoblasts
Alizarin Red S stain
Chondrocytes
Alcian Blue stain
iMSCs
+25 days of diff.
+15 days of diff.
+21 days of diff.
C
iMSCs
Research-Grade BM-MSCs
Clinical-Grade BM-MSCs
Population Doublings
Time (days)

Methods

Ovine model of acute lung injury and sepsis. Sheep were surgically prepared with multiple vascular catheters: femoral and pulmonary (Swan Ganz) arterial, left atrial, and catheter in efferent vessel of mediastinal lymph node draining lung lymph under deep anesthesia and analgesia. After surgical recovery, pneumonia/sepsis was induced by instillation of *Pseudomonas aeruginosa* (~4X10^10 CFU) in to the lungs under anesthesia and analgesia, then sheep were monitored for 48 hrs in a conscious state with mechanical ventilator support.

Treatment and grouping: Two groups of sheep (n=3 in each) were randomized either to *i*-MSCs treatment: 10X10^6 cells/kg intravenously infused at 1 and 24hrs after the injury; or vehicle (PlasmaLite A in same manner).

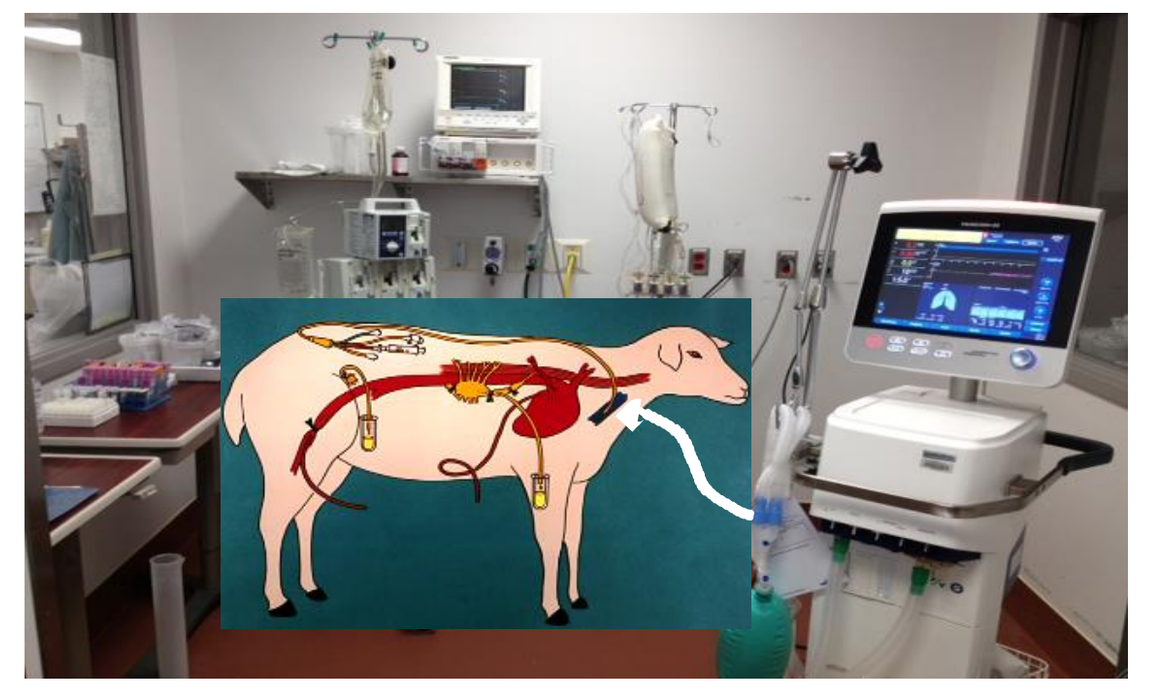

Measured variables:
- Pulmonary gas exchange and
- Pulmonary edema
- Cardiopulmonary hemodynamics
- Fluid balance
- Bacterial clearance

Results

Figure 1. Effects of *i*-MSCs on pulmonary gas exchange (A) and pulmonary shunt fraction (B). *i*-MSCs reduced pulmonary shunt and improved oxygenation, thus preventing onset of ARDS. Data are presented as ±SEM.

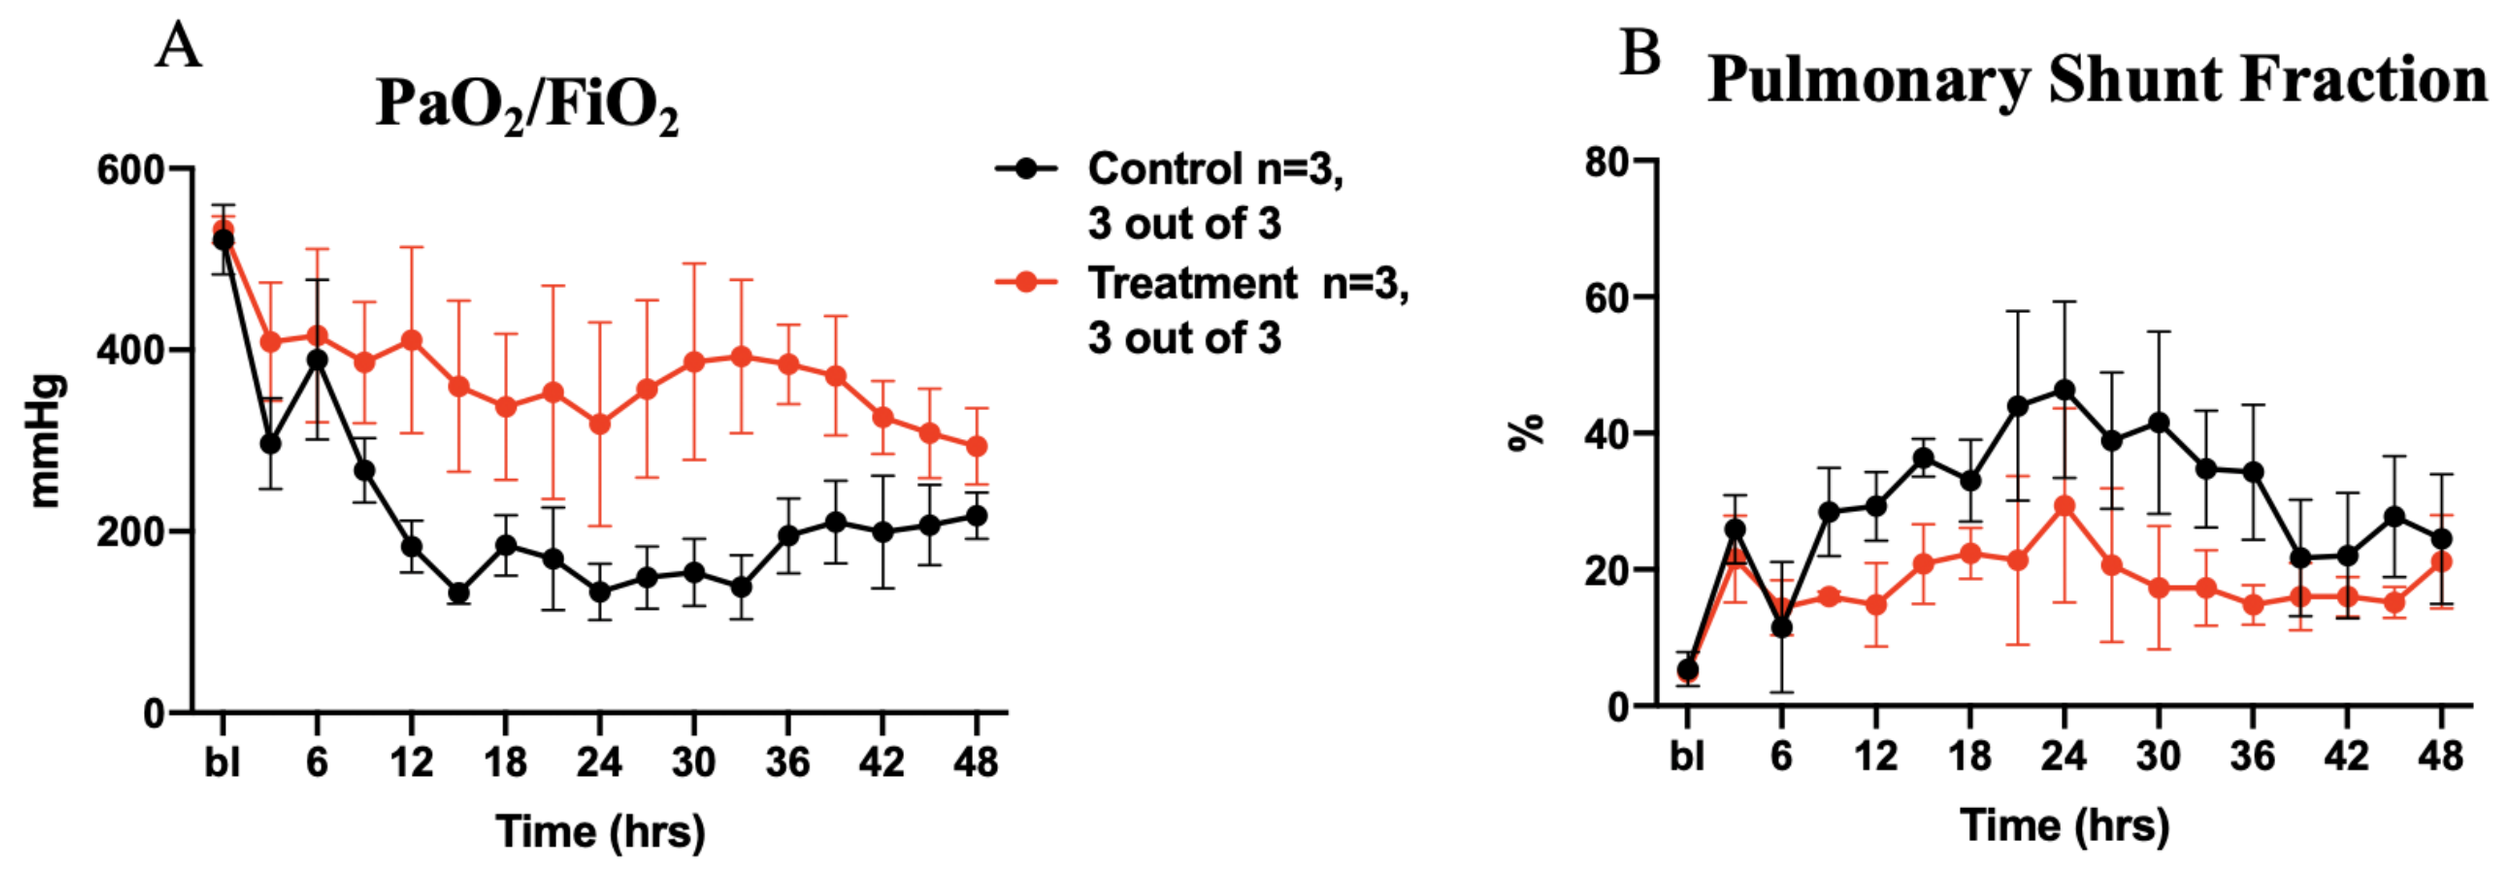

A
PaO2/FiO2
mmHg
Control n=3, 3 out of 3
Treatment n=3, 3 out of 3
Time (hrs)
B
Pulmonary Shunt Fraction
%
Time (hrs)

Figure 2. *i*-MSCs markedly reduced pulmonary edema assessed by measuring lung lymph flow (A) and lung wet-to-dry weight ratio (B). The results indicate *i*-MSCs reduced pulmonary microvascular hyperpermeability to water. Data are expressed as ±SEM.

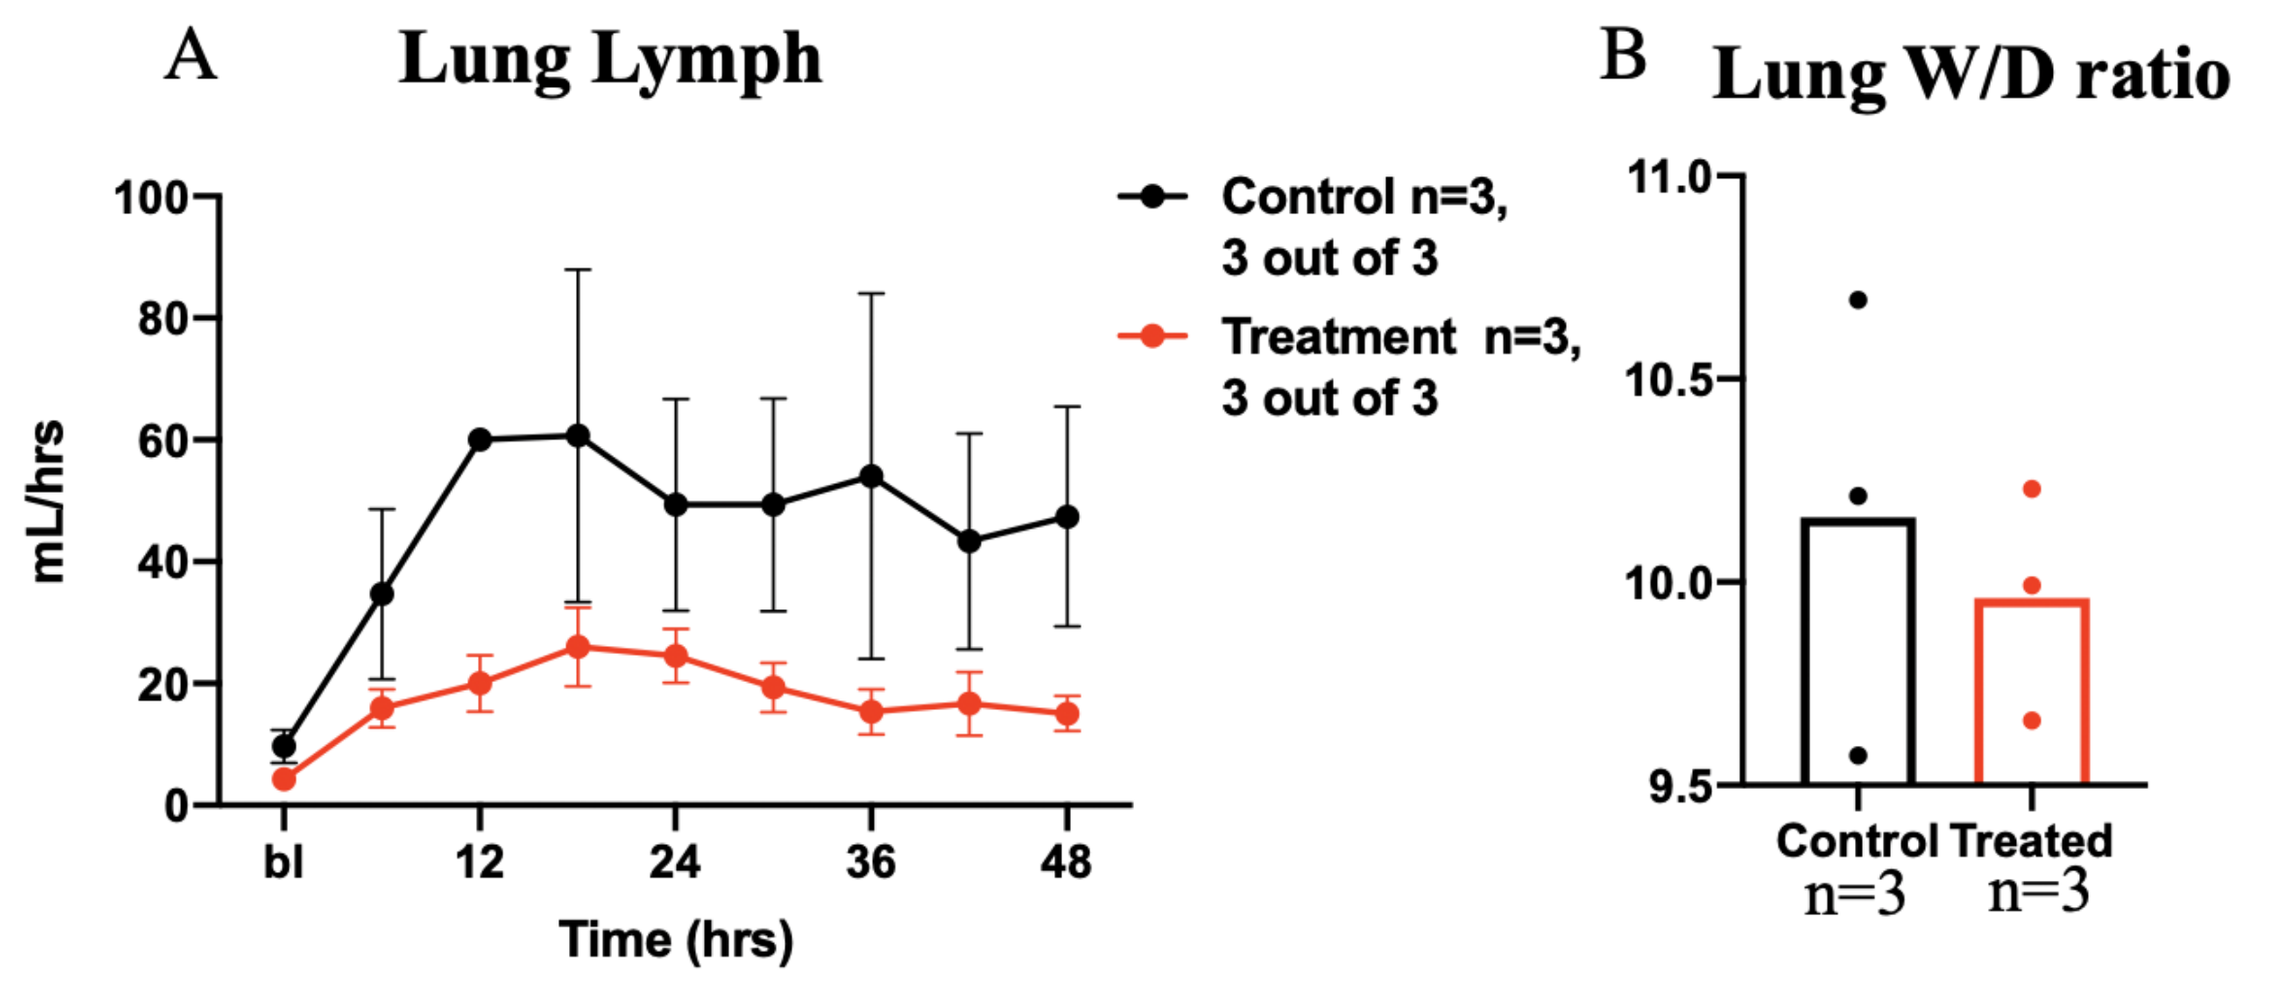

A
Lung Lymph
mL/hrs
Control n=3, 3 out of 3
Treatment n=3, 3 out of 3
Time (hrs)
B
Lung W/D ratio
Control
n=3
Treated
n=3

Results

Figure 3. *i*-MSCs attenuated atelectasis in the lung (A) and better preserved airway epithelial integrity (B). Figures C and D illustrate macroscopic pictures of the lungs and chest X-ray at 48 hrs after the injury.

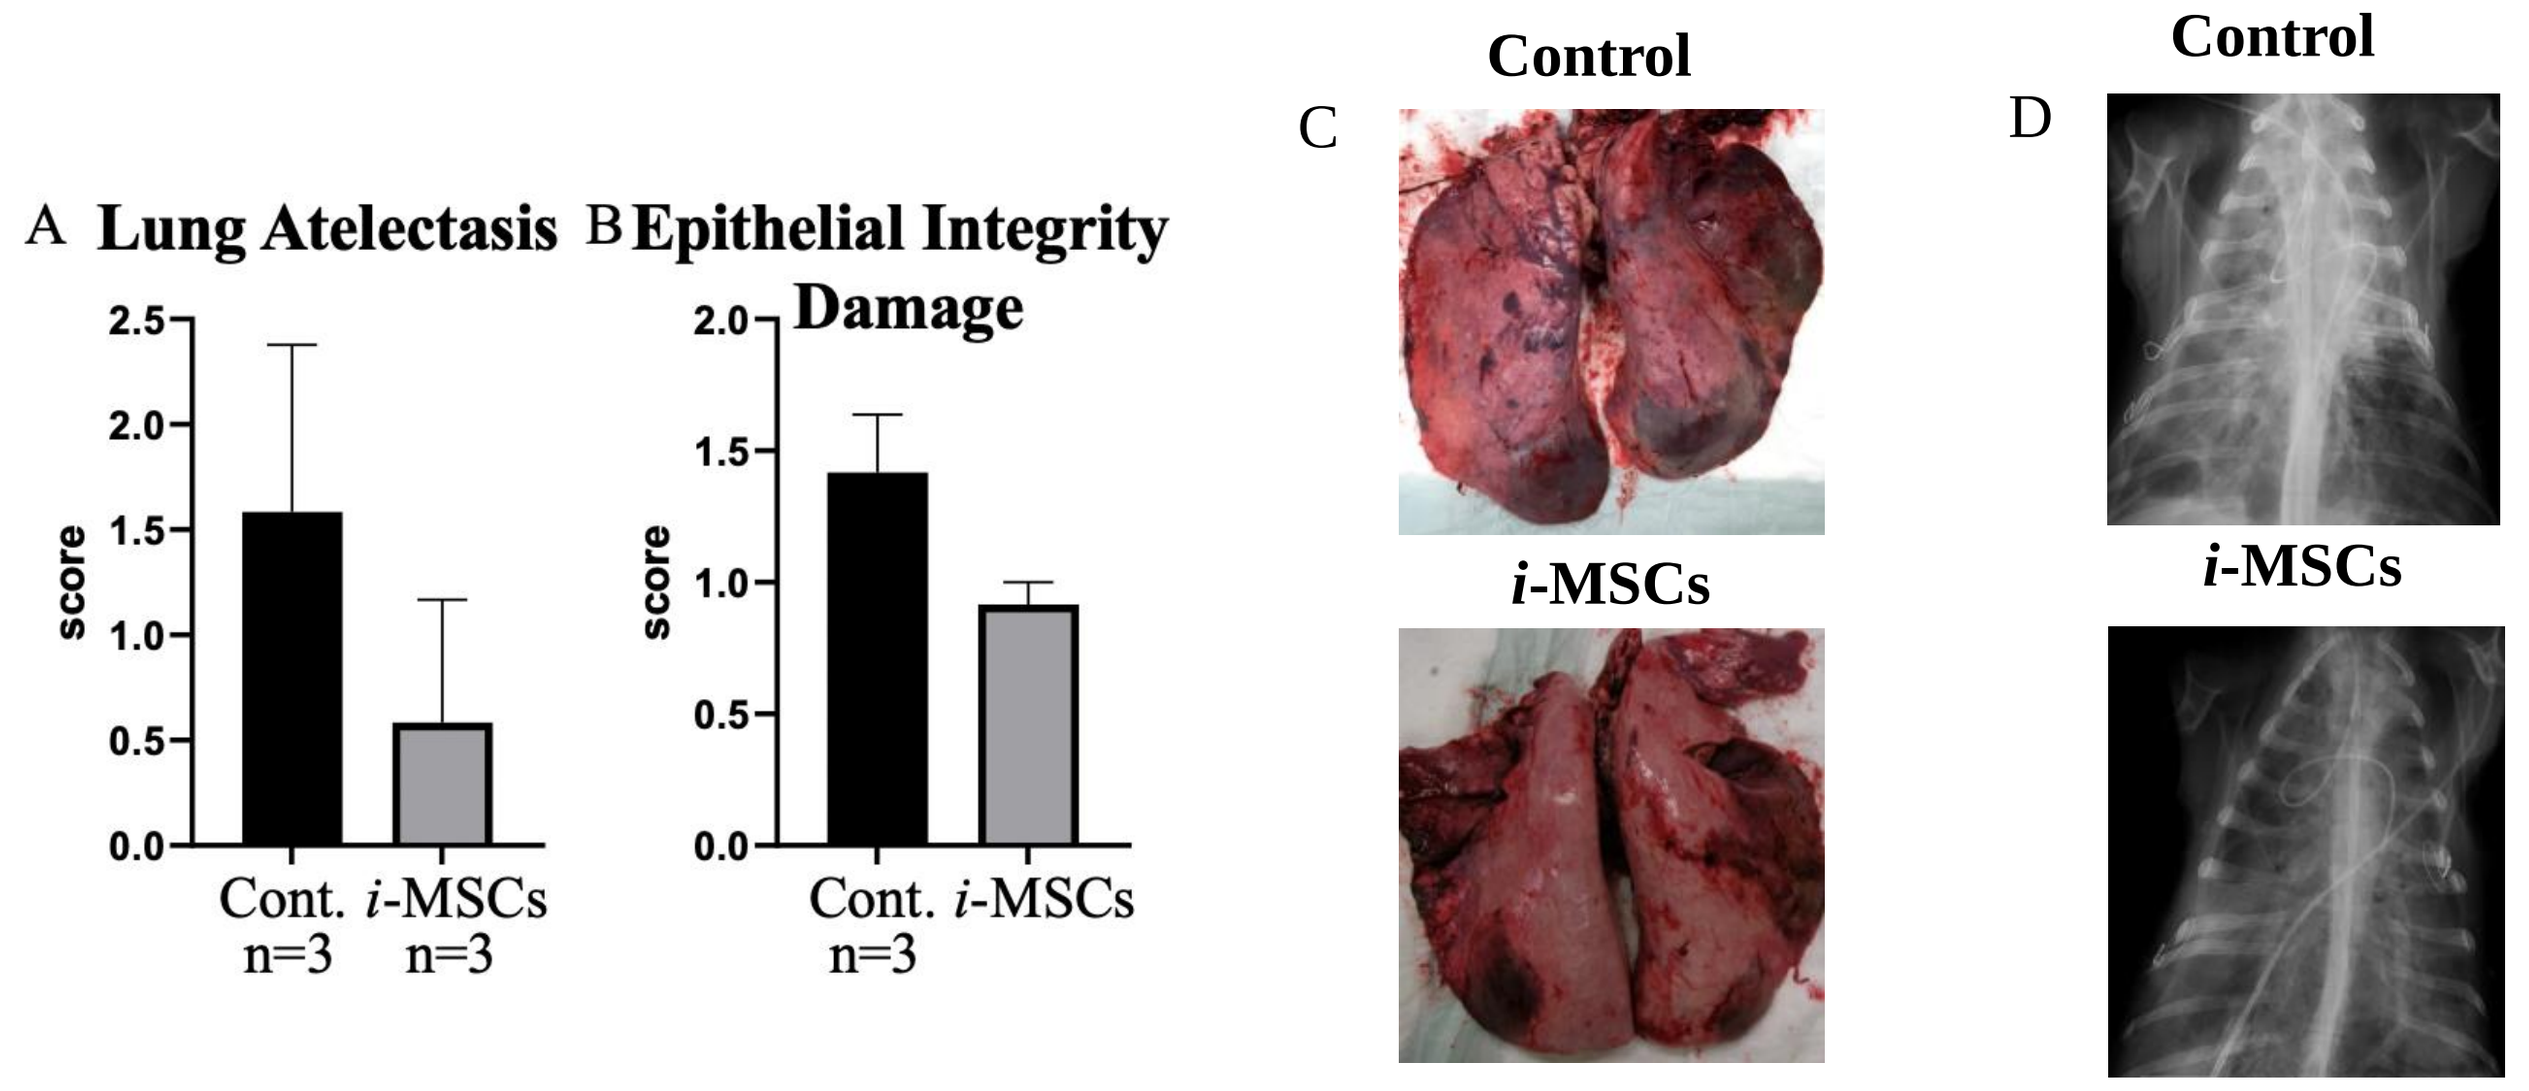

A
Lung Atelectasis
B
Epithelial Integrity Damage
score
Cont.
n=3
i-MSCs
n=3
C
D
Control
i-MSCs

Figure 4. *i*-MSCs reduced pulmonary microvascular hyperpermeability to protein as evidenced by higher plasma (A) and lower lung lymph (B) proteins. Data are expressed as ±SEM.

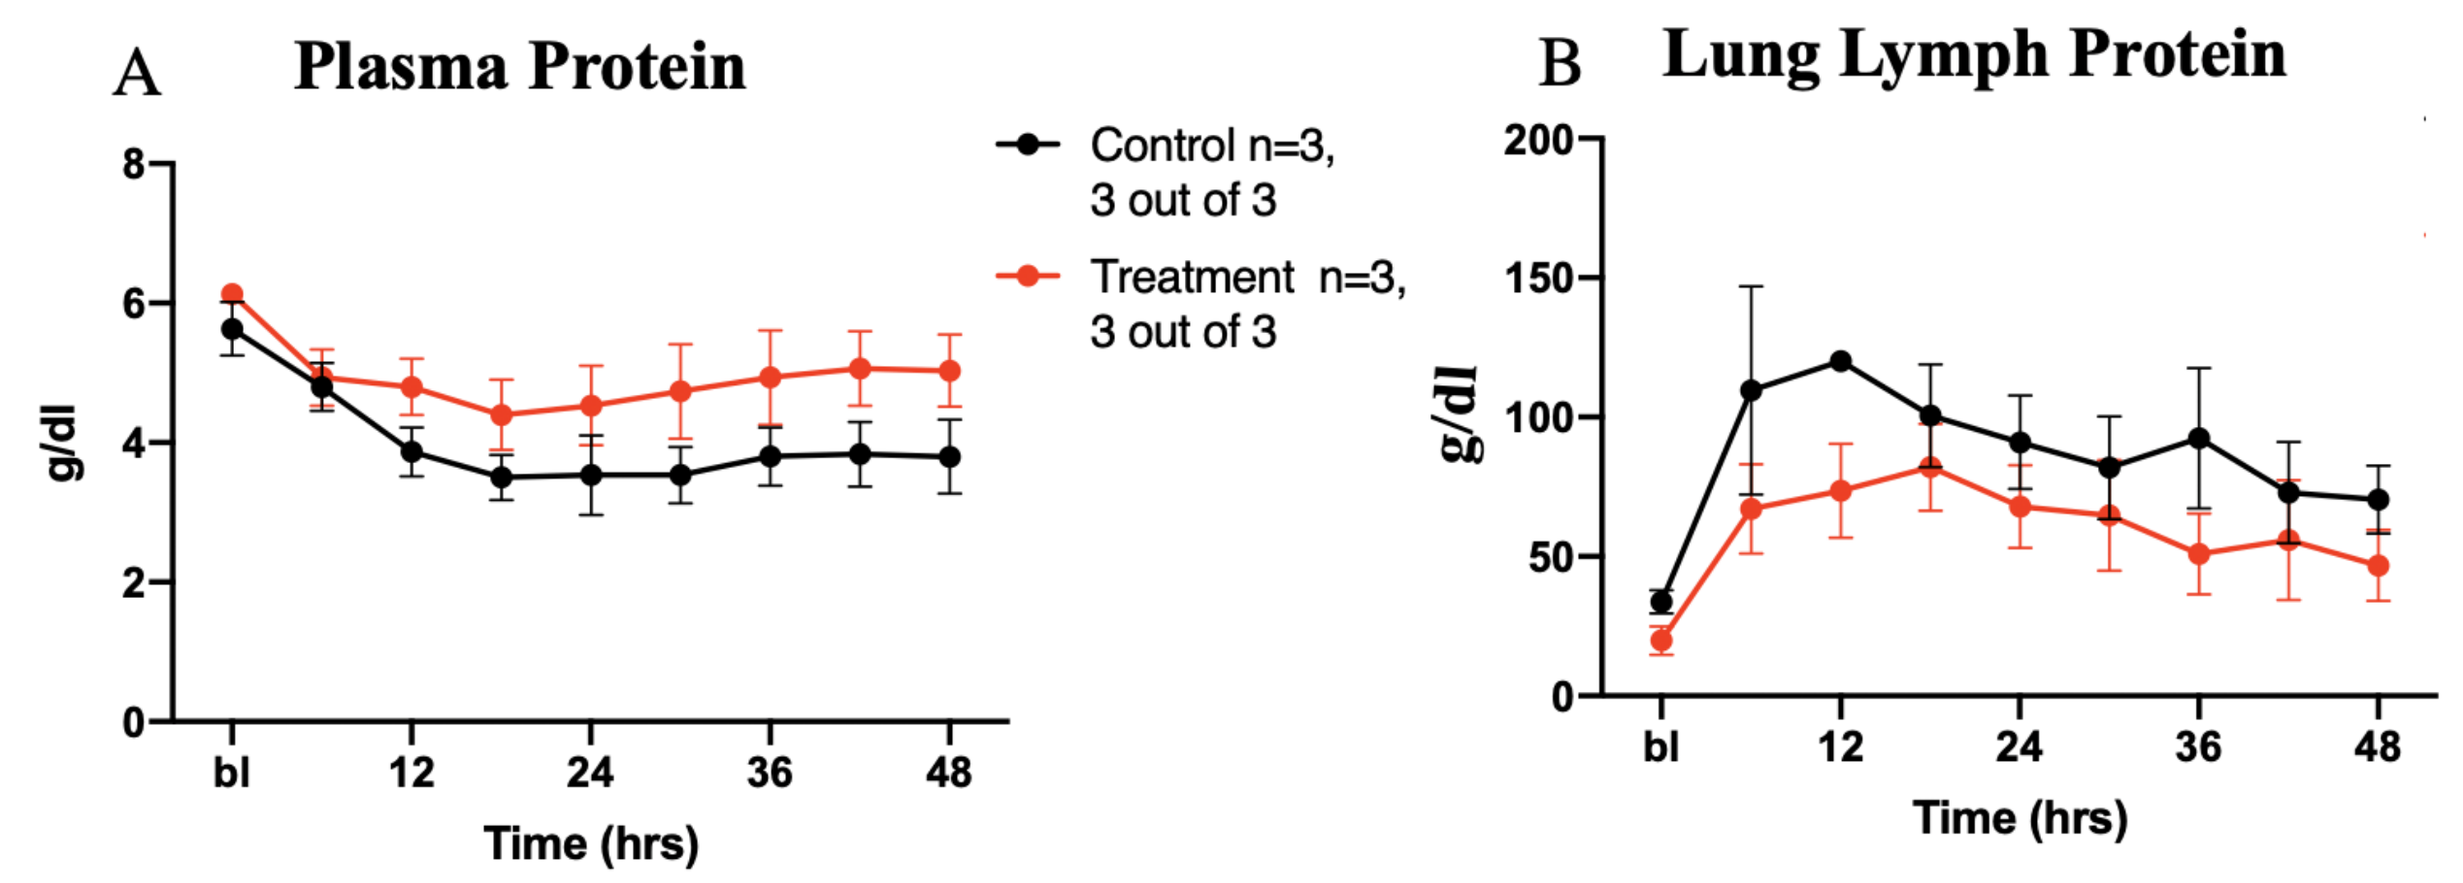

A
Plasma Protein
g/dl
Control n=3, 3 out of 3
Treatment n=3, 3 out of 3
Time (hrs)
B
Lung Lymph Protein
g/dl
Time (hrs)

Figure 5. *i*-MSCs markedly reduced excess fluid requirement (A) and did not augment increases in pulmonary artery pressure (B), indicating that *i*-MSCs are safe to use.

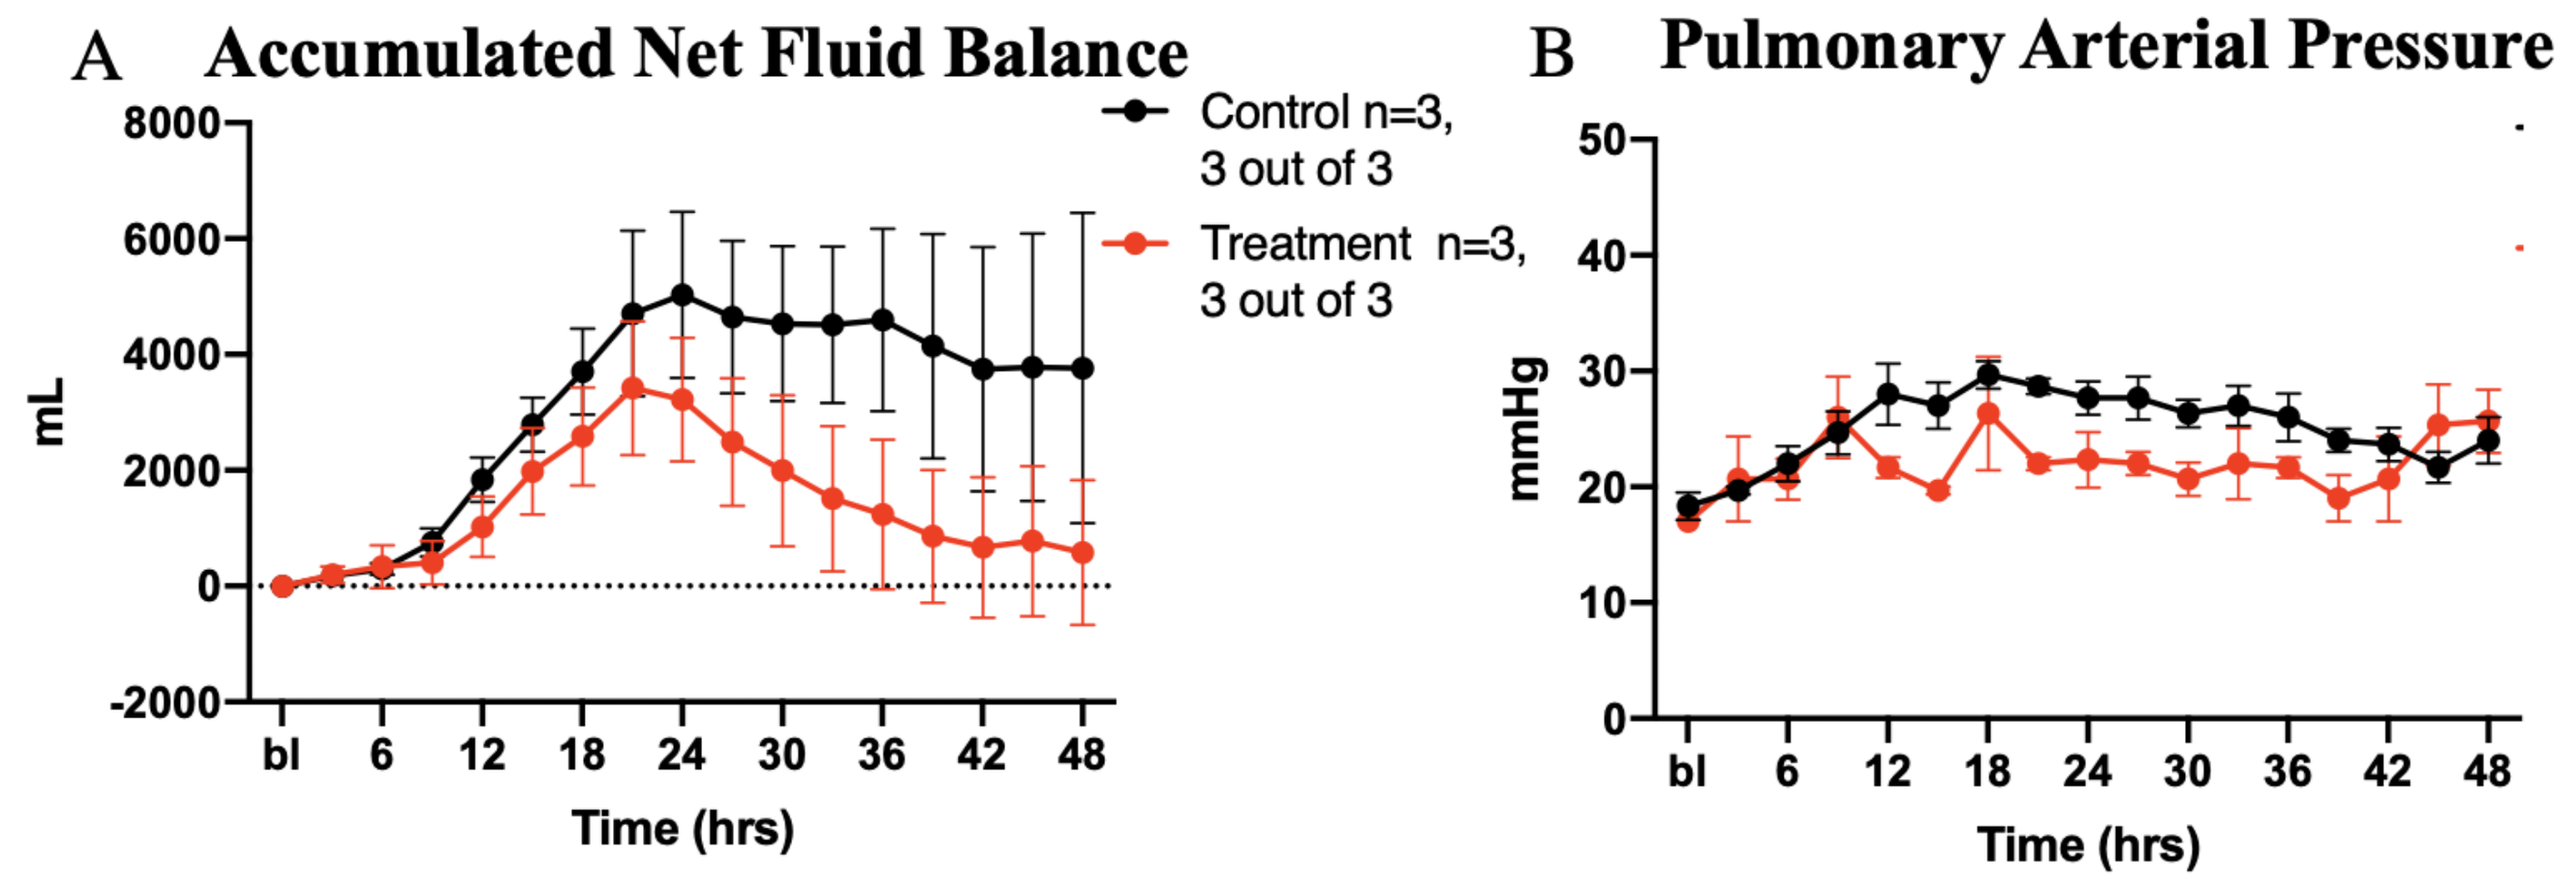

A
Accumulated Net Fluid Balance
mL
Control n=3, 3 out of 3
Treatment n=3, 3 out of 3
Time (hrs)
B
Pulmonary Arterial Pressure
mmHg
Time (hrs)

Figure 6. *i*-MSCs reduced vasopressor (norepinephrine) requirement (B) to maintain arterial blood pressure (A) close to baseline. Data are expressed as ±SEM

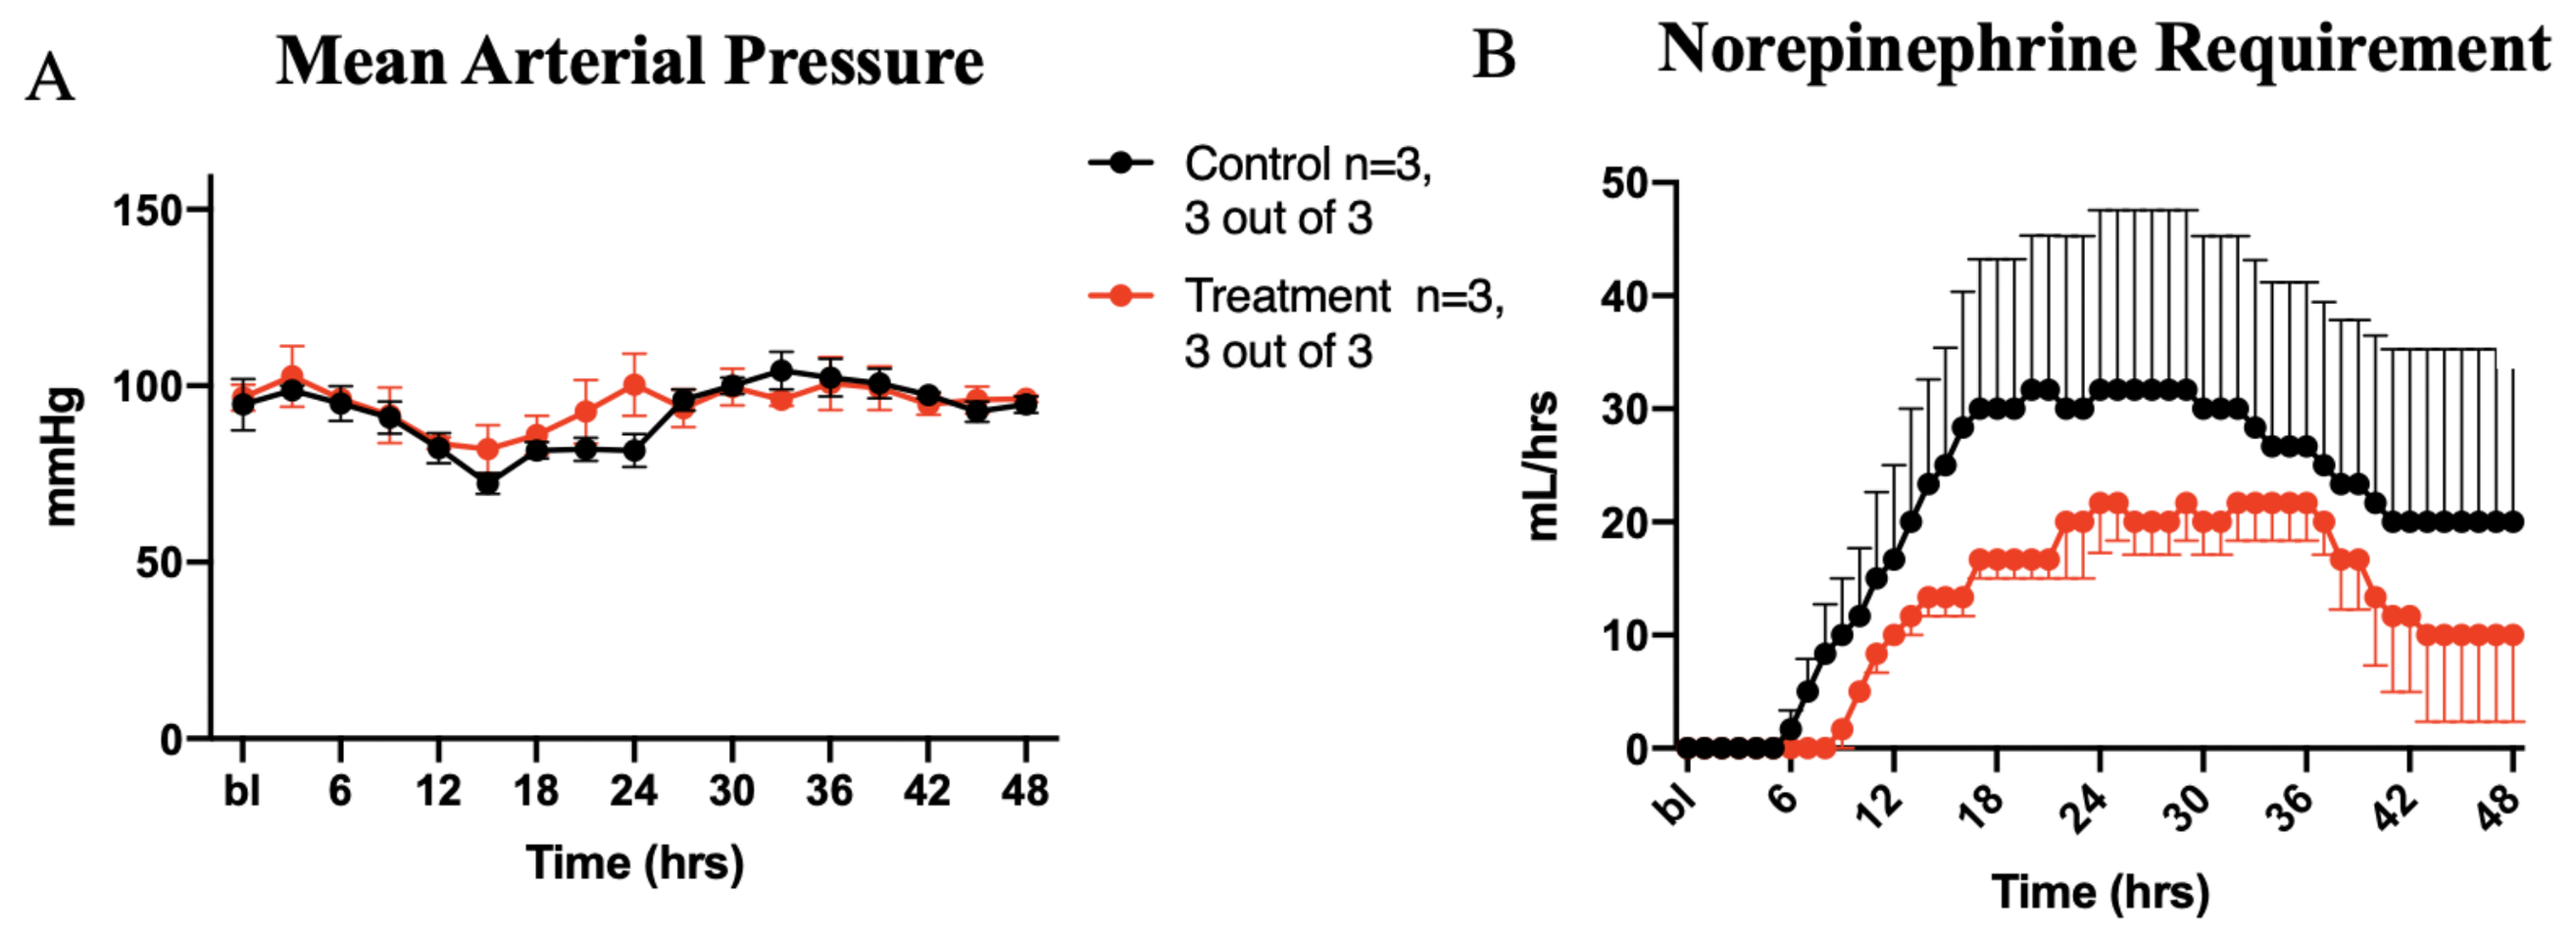

A
Mean Arterial Pressure
mmHg
Control n=3, 3 out of 3
Treatment n=3, 3 out of 3
Time (hrs)
B
Norepinephrine Requirement
mL/hrs
Time (hrs)

Results

Figure 8. *i*-MSCs significantly reduced bacterial burden in lungs, bronchoalveolar lavage fluid (BALF), and spleen (A). Data are expressed as ±SEM. Number of samples in each group is 3. ****p<0.0001; *p<0.05.
B: Macroscopic pictures of bacterial cultures in lung, BALF and spleen (control on bottom; *i*-MSCs on top).

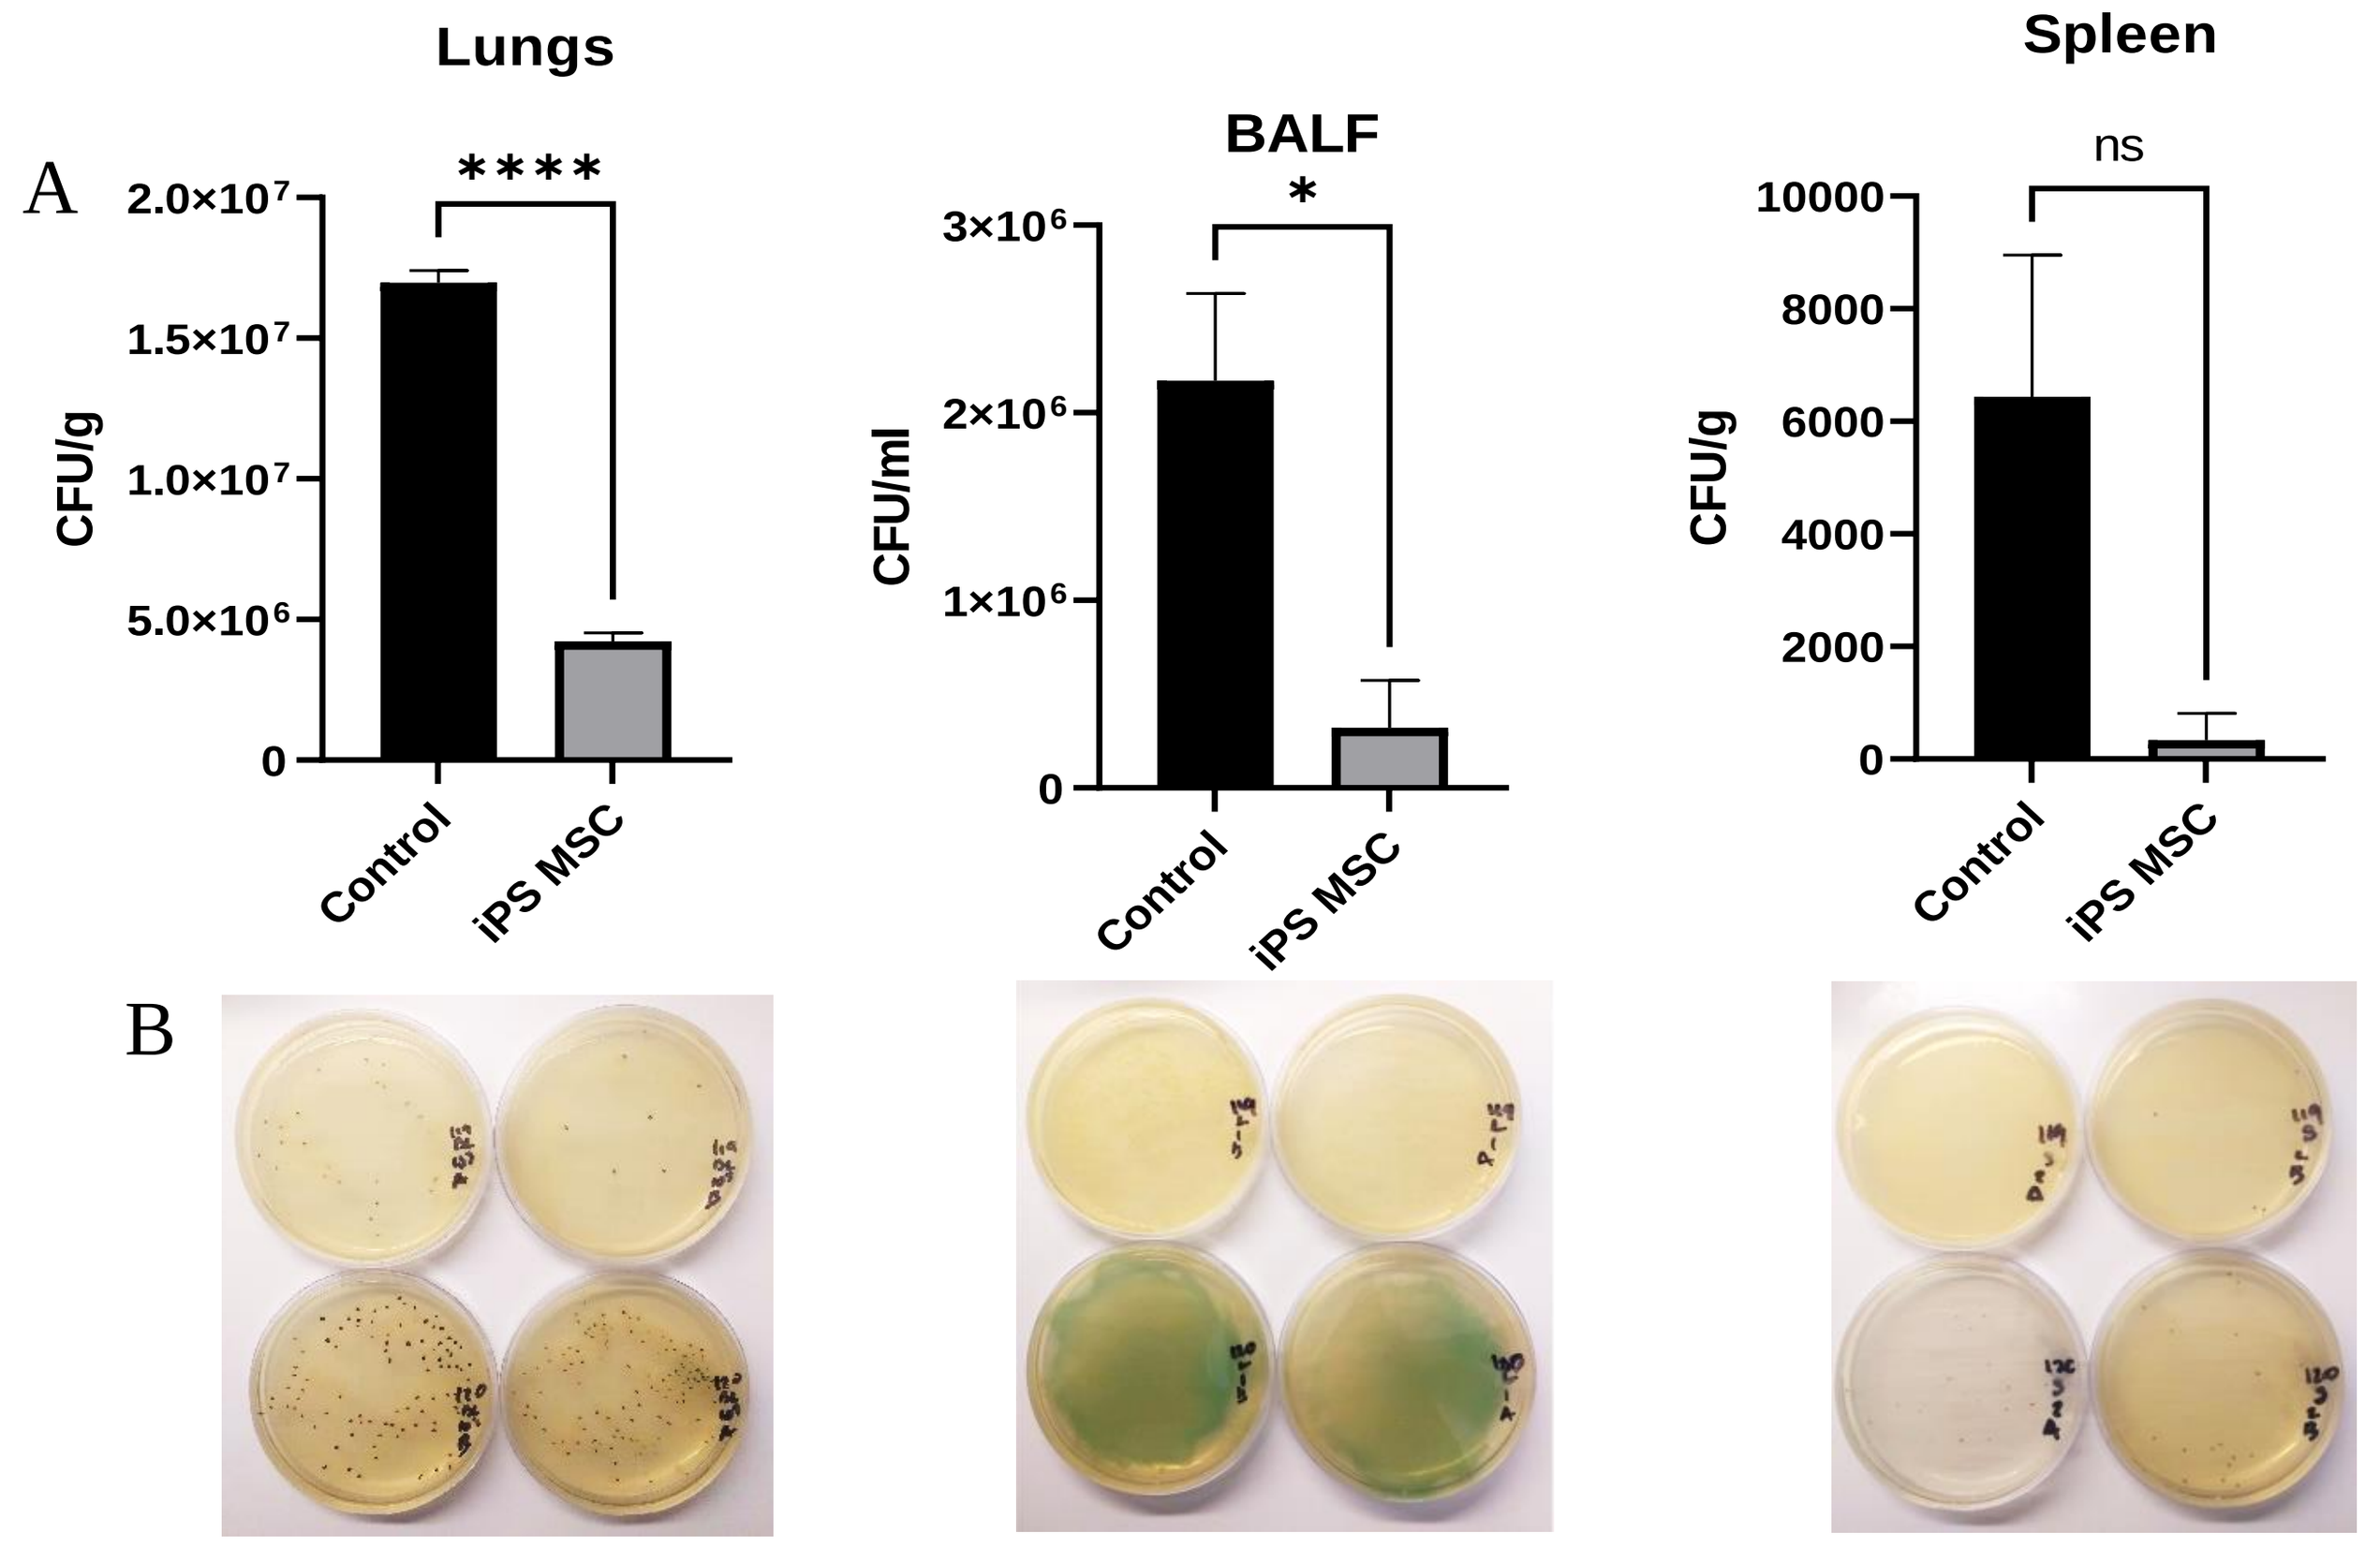

A
Lungs

CFU/g
BALF
*
CFU/ml
Spleen
ns
CFU/g
Control
iPS MSC
B

Conclusions

- *i*-MSCs displayed characteristics of donor MSCs by differentiating to adipocytes, osteoblasts and chondrocytes
- *i*-MSCs population doubling was ~4-fold higher vs. that of bone marrow-derived MSCs
- *i*-MSCs improved oxygenation and prevented onset of ARDS
- *i*-MSCs reduced pulmonary microvascular hyperpermeability to water and protein and ameliorated severity of pulmonary edema
- *i*-MSCs reduced fluid requirement
- *i*-MSCs reduced vasopressor requirement to maintain arterial blood pressure
- *i*-MSCs significantly improved bacterial clearance
- *i*-MSCs had no hemodynamic adverse effects

In summary, use of *i*-MSCs is safe and effective in ameliorating severity of sepsis induced acute lung injury. Further studies increasing sample sizes are warranted. Data from this sheep ARDS study will inform the design and dosing of planned future human clinical trials in ARDS using these *i*-MSCs.

References

1. Matthay MA, et al. *Nat Rev Dis Primers*. 2019, 5:18
2. Asmussen S, et al. *Thorax.* 2014, 69: 819-825
3. Harris J, et al. *Mol Ther,* 2020, 28:4S1

Support: Citius Pharmaceuticals, Inc., Cranford, NJ